Understanding State Medicaid Spending on Prescription Drugs: A 10-State Comprehensive Analysis of Net State Medicaid Drug Spending as Part of the Total State Medicaid Budget

Bridget M. Doherty, MPH, MS[1], Silas Martin, MS[1]

[1]Johnson & Johnson, Titusville, NJ

Background

- In recent years, the cost of healthcare, and specifically prescription drugs, has become a topic of interest for policymakers.
- US State legislatures are considering and advancing legislation on payment limits and other drug pricing policies, including establishing Prescription Drug Affordability Boards (PDABs).
- In 2023, 92 million Americans received drug coverage from Medicaid, a joint federal-state insurance program that provides medical and drug coverage to lower-income Americans. Also in 2023, Medicaid represented about 19%, or around $1 out of every $6, of healthcare spending in the US.
- As the Medicaid program is the source for states to provide health coverage and long-term care services for low-income individuals, Medicaid programs are often under financial pressure and policymakers often focus attention and legislation on drug pricing.

Objective

- Despite widespread stakeholder interest, there is no central source of data to identify the amount that states are spending on prescription drugs as a part of total Medicaid Spending. To put these policy discussions into context, this research aims to provide an estimate of net state Medicaid spending on prescription drugs in 10 states (CA, CO, FL, MA, MI, NY, OH, OR, PA, and WA) in fiscal year (FY) 2023 with significant Medicaid spend and/or policy discussions around spend in FY 2023 based on publicly available sources (e.g., state budgets, Centers for Medicare and Medicaid Services (CMS) reports).

Methods

- State Medicaid drug expenditures and drug rebate income were analyzed to generate an overview of state spending on prescription drugs and estimate Medicaid drug spending as a percentage of total state Medicaid expenditures for 10 states in FY 2023. (See Figure 1)
- Medicaid data sources included:
 - Medicaid State Drug Utilization Data
 - Medicaid Budget and Expenditure System/State Children's Health Insurance Program (CHIP) Budget and Expenditure System Form CMS-64
 - State budget documents

FIGURE 1: Estimating State Medicaid Spending on Drugs

Step 1: Total Medicaid Spending

To determine total Medicaid spending for FY2023, state budget documents and Medicaid websites were reviewed to find the total Medicaid appropriations for the fiscal year.

Step 2: Medicaid State Share of Funding

To estimate the portion of Medicaid prescription drug expenditures paid by the state, the state's assigned FMAP for FY2023 was subtracted from the total Medicaid budget.

Medicaid programs are jointly funded by states and the federal government, with the federal government matching state expenditures.

The Federal Medical Assistance Percentage (FMAP) determines the federal share for most Medicaid expenditures. The state share is 100%-FMAP.

FMAP depends on the state's per-capita income, with the state share lower for states with lower per capita incomes.

FMAP is generally between 50% to 83% of expenditures, but in FY2023 the COVID-19 public health emergency was in effect and Congress temporarily increased FMAP by 6.2 percentage points for states that had expanded Medicaid, making the minimum for the states in this study 56.2%.[i]

50% 56.2% 83%

Step 3: Medicaid Prescription Drug Spending

The net amount that a Medicaid program spends on a particular outpatient prescription drug reflects a payment from Medicaid to the pharmacy or medical provider, and a rebate from the drug manufacturer paid to Medicaid. To estimate state Medicaid expenditures and offsetting rebates, State Drug Utilization Data reports submitted to CMS by states detailing expenditures for outpatient prescribed drugs were reviewed. The FMAP was subtracted from expenditures to determine the state share.

Prescription drugs may be administered in a hospital, provider office, or clinic setting, or may be dispensed by pharmacies for patients to take at home.

All 50 states, D.C., and Puerto Rico cover prescription drugs in Medicaid through the Medicaid Drug Rebate Program (MDRP), which requires manufacturers to enter into an agreement with CMS to provide rebates to state Medicaid programs. In return, states must cover a participating manufacturer's products.

Drugs purchased through the 340B Drug Pricing Program are not eligible for MDRP rebates[iii] and are not included in the utilization data provided to CMS; these expenditures could not be included in the analysis.

Step 4: Medicaid Rebates

To estimate state MDRP rebate collection, reports submitted to CMS by states detailing expenditures and rebates collected were used.[iv] The FMAP was subtracted from rebates collected to determine the state share.

≥23.1%

The rebate for brand drugs is a minimum of 23.1% of the drug's average manufacturer price or more if the manufacturer sells the drug at a steeper discount to certain purchasers, such as a commercial insurance plan. Additional rebates may be applied if the manufacturer has raised the price of the drug greater than the rate of inflation.

>50% reduction

A 2022 Medicaid and CHIP Payment and Access Commission (MACPAC) Report found that MDRP drug rebates reduce gross drug spending by over half.[v]

Step 5: Percentage of State Medicaid Spending

To determine the percentage of state Medicaid spending that is net spending on drugs, the difference in the state share of drug expenditures and rebates was divided by the state share of the total Medicaid budget.

TABLE 1: Calculations to Estimate Gross and Net Drug Spend

Gross (total) Medicaid spending	=	State Medicaid contribution	+	Federal Medicaid contribution
Net state Medicaid spending on drugs	=	State share of Medicaid spending on outpatient, non-340b drugs	–	State share of Medicaid rebates
Percentage of state Medicaid spending that is drug spending	=	Net state Medicaid spending on drugs	÷	State share of Medicaid spending

Step 6: Per-Beneficiary State Drug Spending

As a final step, the net state Medicaid spending on drugs was divided by the number of beneficiaries enrolled in the state's Medicaid program to arrive at the state's per-beneficiary spending on prescription drugs to give further context.

Results

- Using publicly available data, state Medicaid net prescription drug spending as a percentage of overall state Medicaid budget ranged from 4.7% to 14.3% across 10 states. These findings are near or below the national rate of drug spending as a share of overall healthcare spending, 14%, which is also below the average drug spending share of medical spending across comparable countries.[vi,vii] Manufacturer drug rebates back to states ranged from $83M (OR) to $1.9B (NY).

TABLE 2: Summary of State Medicaid Spending, FY2023

State	Total Medicaid Budget	Medicaid Budget, State Share	Gross (Pre-Rebate) Spending on Drugs, State Share	Drug Rebate Income, State Share	Net Medicaid Spending on Drugs, State Share	Percentage of State Medicaid Spending on Prescription Drugs, Net of Rebates
CA	$144.2B	$54.9B	$5.7B	$1.3B	$4.3B	7.9%
CO	$13.5B	$4.8B	$673M	$240M	$433M	9.0%
FL	$36.4B	$12.3B	$1.3B	$716M	$582M	4.7%
MA	$18.6B	$7.2B	$971M	$407M	$563M	7.9%
MI	$24.3B	$6.1B	$1.1B	$414M	$662M	10.8%
NY	$92.0B	$28.7B	$3.7B	$1.9B	$1.9B	6.5%
OH	$36.1B	$9.6B	$1.2B	$561M	$648M	6.7%
OR	$13.2B	$3.8B	$281M	$83M	$198M	5.2%
PA	$28.3B	$9.7B	$1.8B	$687M	$1.1B	11.5%
WA	$9.7B	$3.0B	$696M	$256M	$440M	14.3%

FIGURE 2: Annual State Medicaid Drug Spending Per Beneficiary, FY 2023

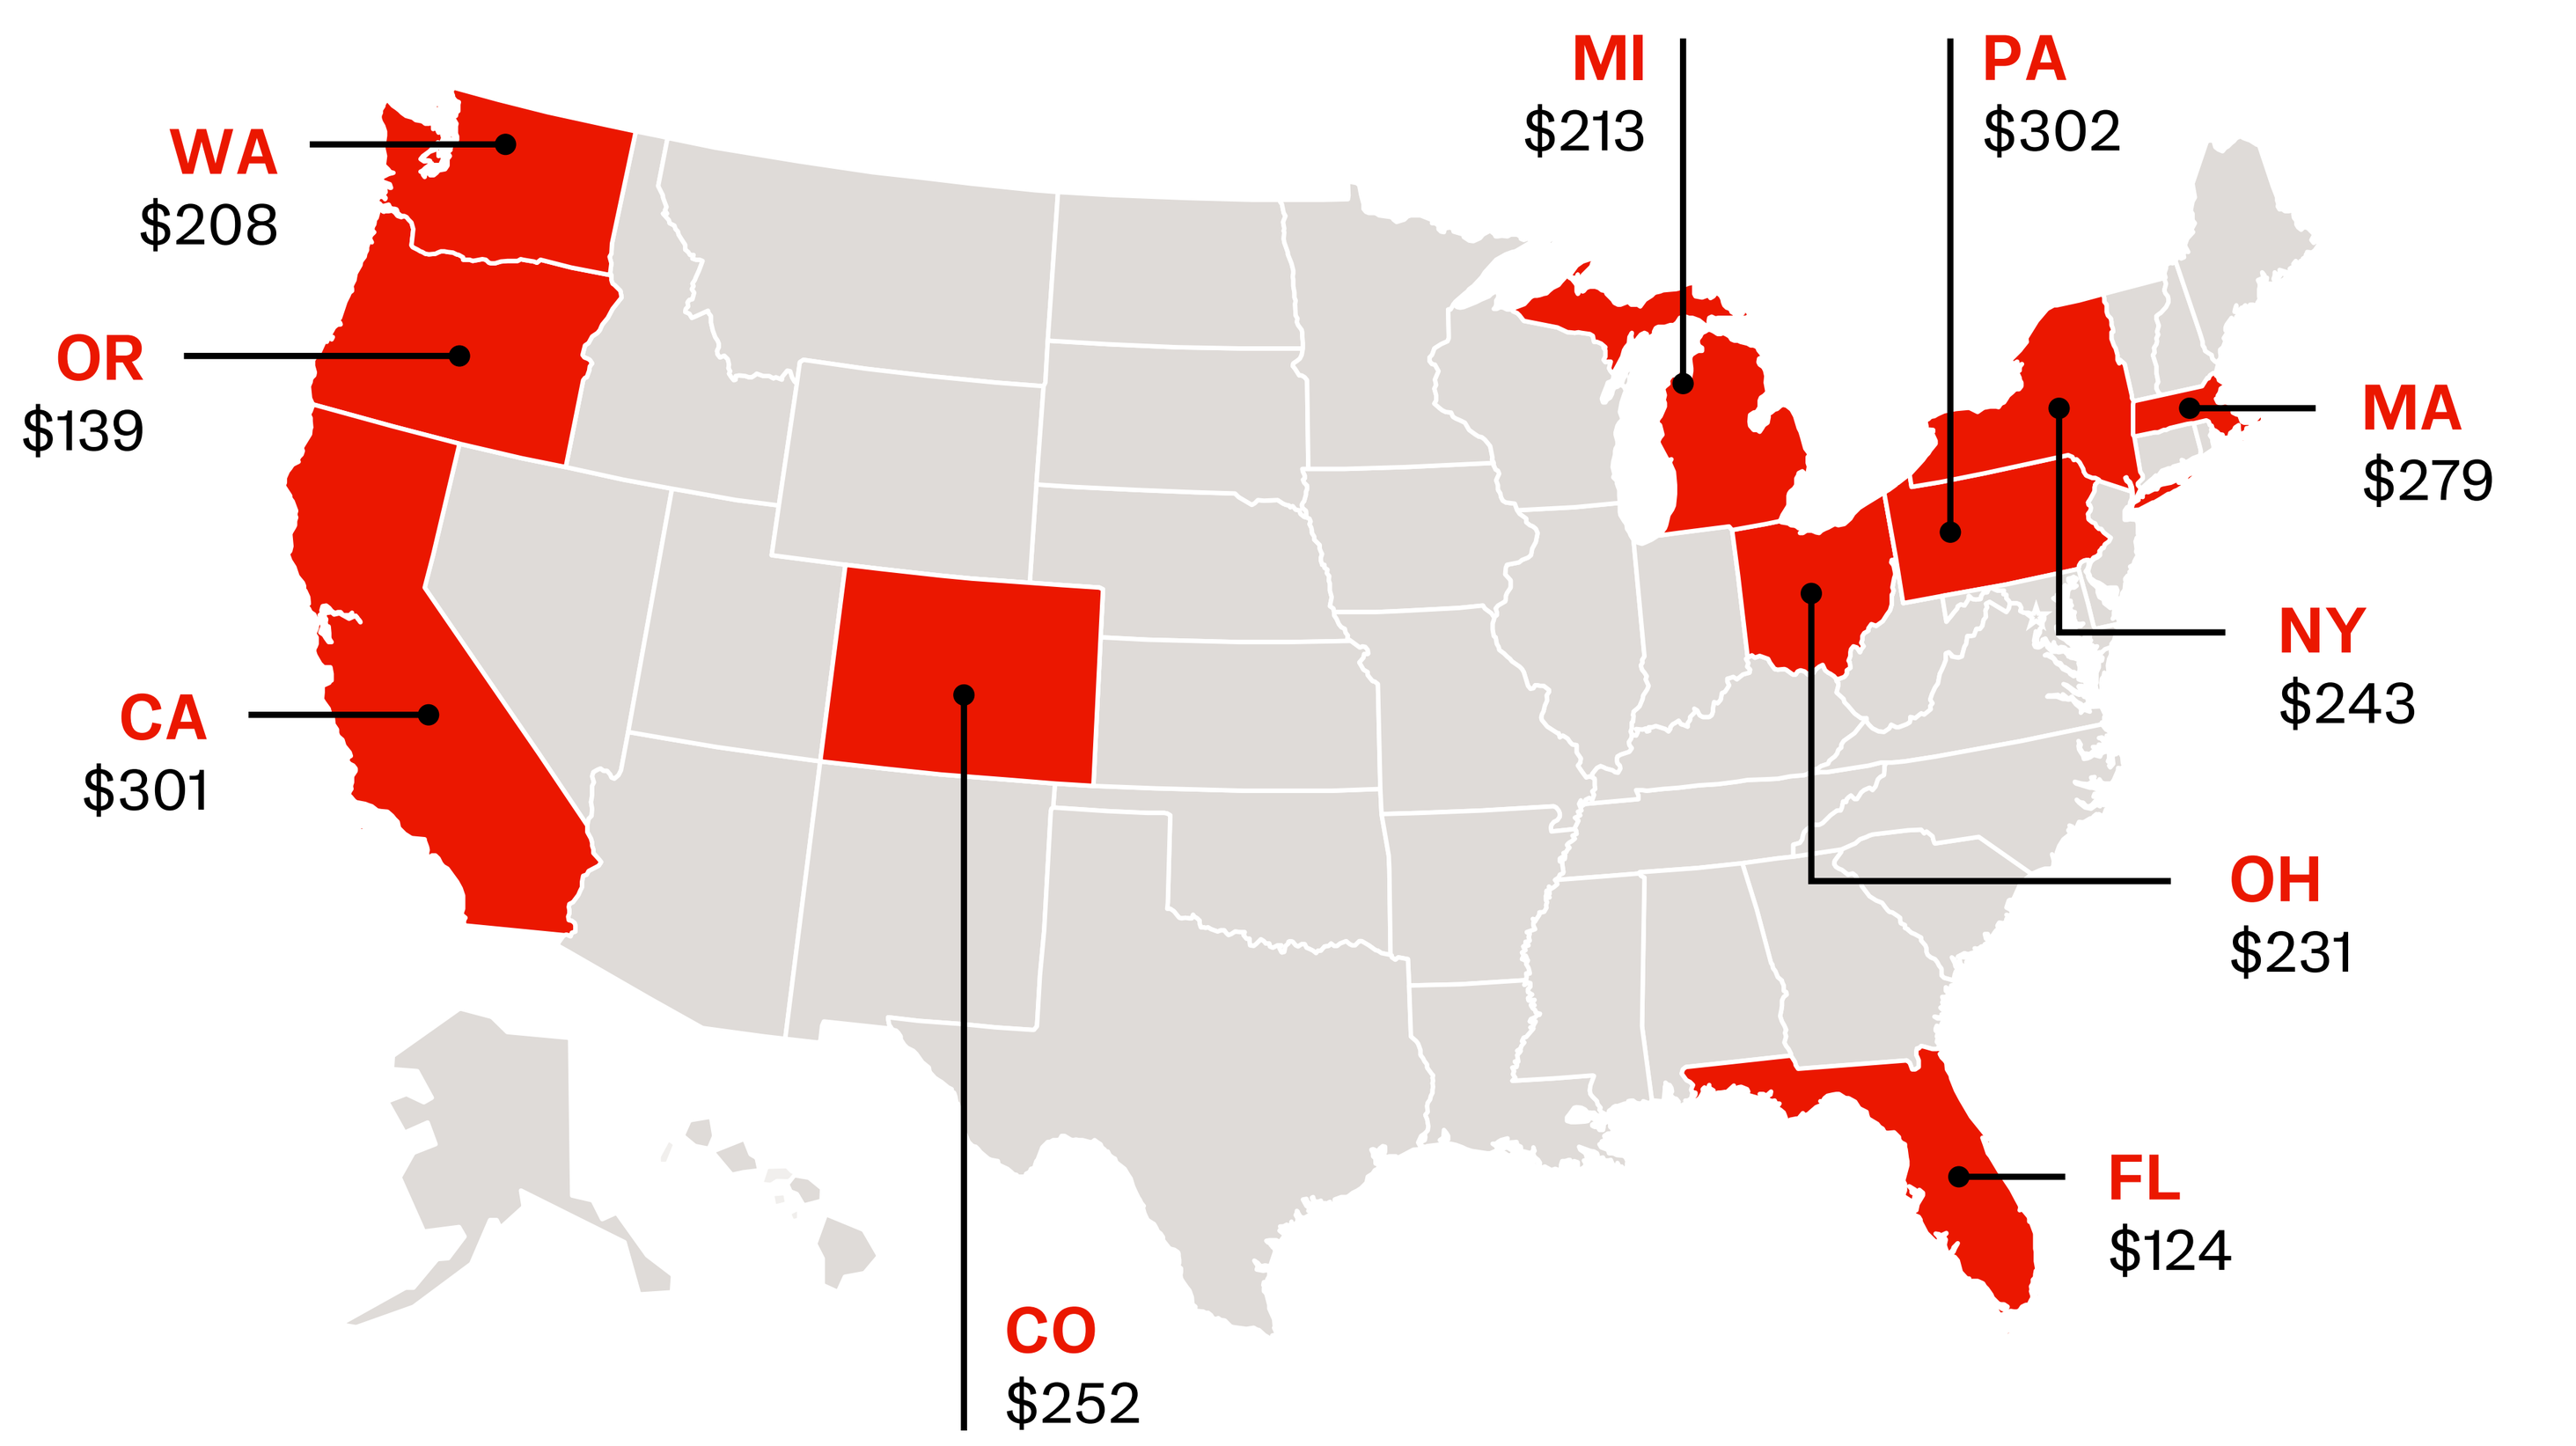
WA
$208
OR
$139
CA
$301
CO
$252
MI
$213
PA
$302
MA
$279
NY
$243
OH
$231
FL
$124

Limitations

Factors that impact state spending on prescription drugs and the extent of transparency in state reporting on drug spending were identified:

- **Uncertainty regarding inpatient hospital spending:** The cost of inpatient drugs was not included. Drugs used for hospital inpatients are generally bundled into a diagnosis related group or other prospective bundled payment where drug costs are not separately identified. These drugs are not eligible for MDRP rebates and are not reported by states.
- **State Share of Fee for Service vs. Managed Care Enrollees:** Medicaid beneficiaries may be covered under a state's fee for service (FFS) Medicaid program, in which the state manages the drug benefit and bears the financial responsibility for drug costs, or in a managed care organization (MCO) arrangement, in which the state contracts with an MCO to administer Medicaid benefits.
 - FFS models generally offer more transparency in drug pricing compared to MCOs. States that carve out pharmacy benefits from MCOs (like CA, NY, OH) may have greater control and transparency over pharmacy benefit spending than provider-administered drug costs.
 - Within managed care, MCOs cover and pay for medical care. States can either "carve in" or "carve out" drug benefit management:
 - **Carve in:** Prescription drug coverage and risk are included in MCO contracts and therefore difficult to assess within the total cost. Some states implement a unified preferred drug list (PDL), requiring MCOs to follow the state's established PDL for coverage, even if the drug benefit is included in the MCO contract.
 - **Carve out:** The state manages the drug benefit separately and bears the financial responsibility. Because they have a more direct line of sight to medication spend, states that carve the pharmacy benefit out of MCOs (CA, NY, OH) may have more transparency and control over pharmacy benefit spending than provider-administered drug spending, which can be wrapped into other line items
- **340B Drug Spend:** 340B drug spending is excluded from state drug utilization data, likely leading to underestimates of total Medicaid drug spending. Much of the reporting on drug spending per state is related to MDRP. Drugs purchased through 340B are not eligible for MDRP rebates, therefore it is hard to quantify how much spending by state Medicaid agencies is on 340B drugs, and whether these drugs cost more or less than the net cost of non-340B drugs after rebates.
- **FMAP Variation:** The FMAP statutory minimum is set at 50% and the statutory maximum at 83%. FMAP varies considerably across states and is dependent upon key factors related to the state's economy, (e.g., per-capita personal income in relation to the US average), state Medicaid expansion status, and enhanced FMAP for certain populations, providers, and services (e.g., family planning).
- **Lack of transparency:** States vary as to what types of information is published relating to Medicaid spending.

Conclusions

- This analysis demonstrates that state Medicaid programs are estimated to be spending near, or below, the US national average on prescription drugs.
- While high drug pricing makes headlines and is often the focus of legislation and policymaker attention, drug pricing is not the main driver of state Medicaid spending based on this estimate.
- Discussions around healthcare spending and drug pricing should be rooted in a factual understanding of what states are spending on medications and the value that spending may bring, as well as a better understanding of what is driving healthcare costs in individual states to help this conversation and drive better policy solutions.

**Presented at AMCP Nexus 2025;
October 27–30, 2025; National Harbor, Maryland, USA**

Scan the QR code

The QR code is intended to provide scientific information for individual reference, and the information should not be altered or reproduced in any way.

Poster Number 135

REFERENCES:

i. Medicaid and CHIP Payment and Access Commission (2024), EXHIBIT 6. Federal Medical Assistance Percentages (FMAPs) and Enhanced FMAPs (E-FMAPs) by State. https://www.macpac.gov/wp-content/uploads/2024/12/EXHIBIT-6.-FMAP-and-Enhanced-FMAP-by-State-FYs-2022%E2%80%932025.pdf. **ii.** Medicaid.gov (2025), State Drug Utilization Data. https://www.medicaid.gov/medicaid/prescription-drugs/state-drug-utilization-data. **iii.** Health Resources and Services Administration (2020), 340B Drug Pricing Program Duplicate Discount Prohibition. https://www.hrsa.gov/opa/program-requirements/medicaid-exclusion. **iv.** Medicaid.gov (2023), Expenditure Reports. Medicaid Budget and Expenditure System/State Children's Health Insurance Program (CHIP) Budget and Expenditure System (MBES/CBES) Form CMS-64. https://www.medicaid.gov/medicaid/financial-management/state-expenditure-reporting-for-medicaid-chip/expenditure-reports-mbescbes. **v.** Medicaid and CHIP Payment and Access Commission (2022), Trends in Medicaid Drug Spending and Rebates. https://www.macpac.gov/wp-content/uploads/2022/10/07_Trends-in-Medicaid-Drug-Spending-and-Rebates-Chris.pdf. **vi.** IQVIA (2021) Drug Expenditure Dynamics 1995-2020. https://www.iqvia.com/insights/the-iqvia-institute/reports-and-publications/reports/drug-expenditure-dynamics. This data point is from the appendix: https://www.iqvia.com/-/media/iqvia/pdfs/institute-reports/drug-expenditure-dynamics/appendices/drug-expenditure-analysis_countryslides_us_v01.pdf. **vii.** OECD. Health at a Glance 2023. https://www.oecd.org/content/dam/oecd/en/publications/reports/2023/11/health-at-a-glance-2023_e04f8239/7a7afb35-en.pdf.

This material is distributed for scientific purposes on J&J Medical Connect, and is not for promotional use