Value-based care: Insights from a survey of patients with cancer in the United States

Rebekah SM Angove[1], Kathleen D Gallagher[1], Christine Brittle[2], Kimberly D Brunisholz[3], Alan Balch[1]

[1]Patient Advocate Foundation, Hampton, VA, USA; [2]Uncommon Insights, LLC, Arlington, VA, USA; [3]Johnson & Johnson, Titusville, NJ, USA

Introduction

- Value-based care (VBC) and value-based payment (VBP) models are increasingly used within cancer care to measure quality, promote cost containment, and guide provider reimbursement based on defined metrics of quality[1,2]
 - An example VBP model in oncology is Medicare's Enhancing Oncology Model[3]
- However, there is limited literature on VBC and aspects of quality in care from the perspective of patients living with cancer[1]

Methods

Study design and population

- This cross-sectional survey was designed to understand patient insights on VBC in cancer care
 - It was conducted by the Patient Advocate Foundation from October to November 2025 and was delivered online via Qualtrics
 - It consisted of 40 items, including two open-ended questions
 - Prior to deployment, it was pilot-tested using virtual cognitive interviews with 10 participants who were representative of the target study sample
 - Digital gift cards worth US$25 were awarded at random to three survey respondents as an incentive to take part
- The survey was distributed to invitees who had previously signed up to receive survey invitations from the Patient Advocate Foundation
 - Eligibility criteria included being ≥19 years of age, living in the United States, having a chronic health condition, and speaking English

Analyses

- The data presented here are from the subset of respondents with cancer and have been analyzed categorically using frequencies and percentages
 - Two-tailed significance testing was used to evaluate whether outcomes differed by cancer risk level, care setting, gender, sexual orientation, age, race, education, insurance, or geographic setting
- Open-ended questions were coded using inductive thematic analysis to establish high-level themes and response categories

Results

Study population

- Overall, there were 830 survey respondents in the cancer subset, representing more than 30 different cancer types
- The majority were female (n=521 [63%]), White (n=577 [70%]), ≥56 years old (n=629 [76%]), had a total household income of less than US$48,000 (n=474 [57%]), had not completed a full college-level education (n=459 [55%]), and had only Medicare insurance (n=496 [60%]) (**Table 1**)

TABLE 1: Respondent demographics and clinical characteristics

Characteristic	Survey respondents (n=830)	Characteristic	Survey respondents (n=830)
Gender		**Annual household income (USD) in the past 12 months**	
Male	309 (37)	<$23,999	186 (22)
Female	521 (63)	$24,000 to $47,999	288 (35)
Age category, years		$48,000 to $71,999	125 (15)
19–35	9 (1)	$72,000 to $95,999	62 (7)
36–55	192 (23)	$96,000 to $119,999	34 (4)
56–75	475 (57)	≥$120,000	23 (3)
≥75	154 (19)	Prefer not to disclose	112 (13)
Race/ethnicity		**Primary insurance type**	
White	577 (70)	COBRA continuation coverage	6 (1)
Black/African American	175 (21)	Employer-based	121 (15)
Hispanic/Latino	70 (8)	Marketplace Exchange	44 (5)
Asian	18 (2)	Medicaid	74 (9)
American Indian/Alaska Native	16 (2)	Medicare	496 (60)
Other[a]	5 (1)	Dual eligible (Medicaid and Medicare)	42 (5)
Highest educational attainment		Other	34 (4)
Lower than high school	11 (1)	No insurance	13 (2)
High school or GED	157 (19)	**Cancer type[b]**	
Some college or 2-year degree	268 (32)	Multiple myeloma	249 (30)
Other degree or certificate	23 (3)	Breast	236 (28)
College degree	207 (25)	Prostate	90 (11)
Master's degree	119 (14)	Lung	47 (6)
Graduate or professional degree	45 (5)	Colon or rectal	44 (5)
Care setting		Other	399 (48)
Community-based care	250 (30)		
Academic medical center	358 (43)		
Both community-based care and academic medical center	168 (20)		
Not sure	50 (6)		

Data are n (%). Percentages are rounded and may not add up to 100. [a]Includes Middle Eastern or North African (n=4) and Native Hawaiian or Pacific Islander (n=1). [b]Multiple responses were possible; therefore, the total exceeds n=830. **COBRA**, Consolidated Omnibus Budget Reconciliation Act; **GED**, General Educational Development; **USD**, US dollars.

Familiarity with VBC

- Respondents had limited awareness of and familiarity with VBC, with 70% having never heard of it before and only 9% recalling their providers having discussed it with them (**Figure 1**)
 - Awareness and familiarity were consistent across cancer risk level, care setting, gender, age, race, education, insurance, and geographic setting (data not shown)
 - Non-White respondents, those with lower educational attainment, and Medicaid-insured or dual-enrolled respondents were more likely than others to report both provider participation in a VBP model and discussions with providers about VBC (data not shown)

Perceptions of VBC, quality in care, and cost containment

- Almost half of respondents (48%) were concerned, 44% were unsure, and 8% were excited about VBC (**Table 2**)
 - Although respondents trusted their providers, they felt that cost incentives may decrease the quality of care

FIGURE 1: Awareness of and familiarity with VBC (n=830)

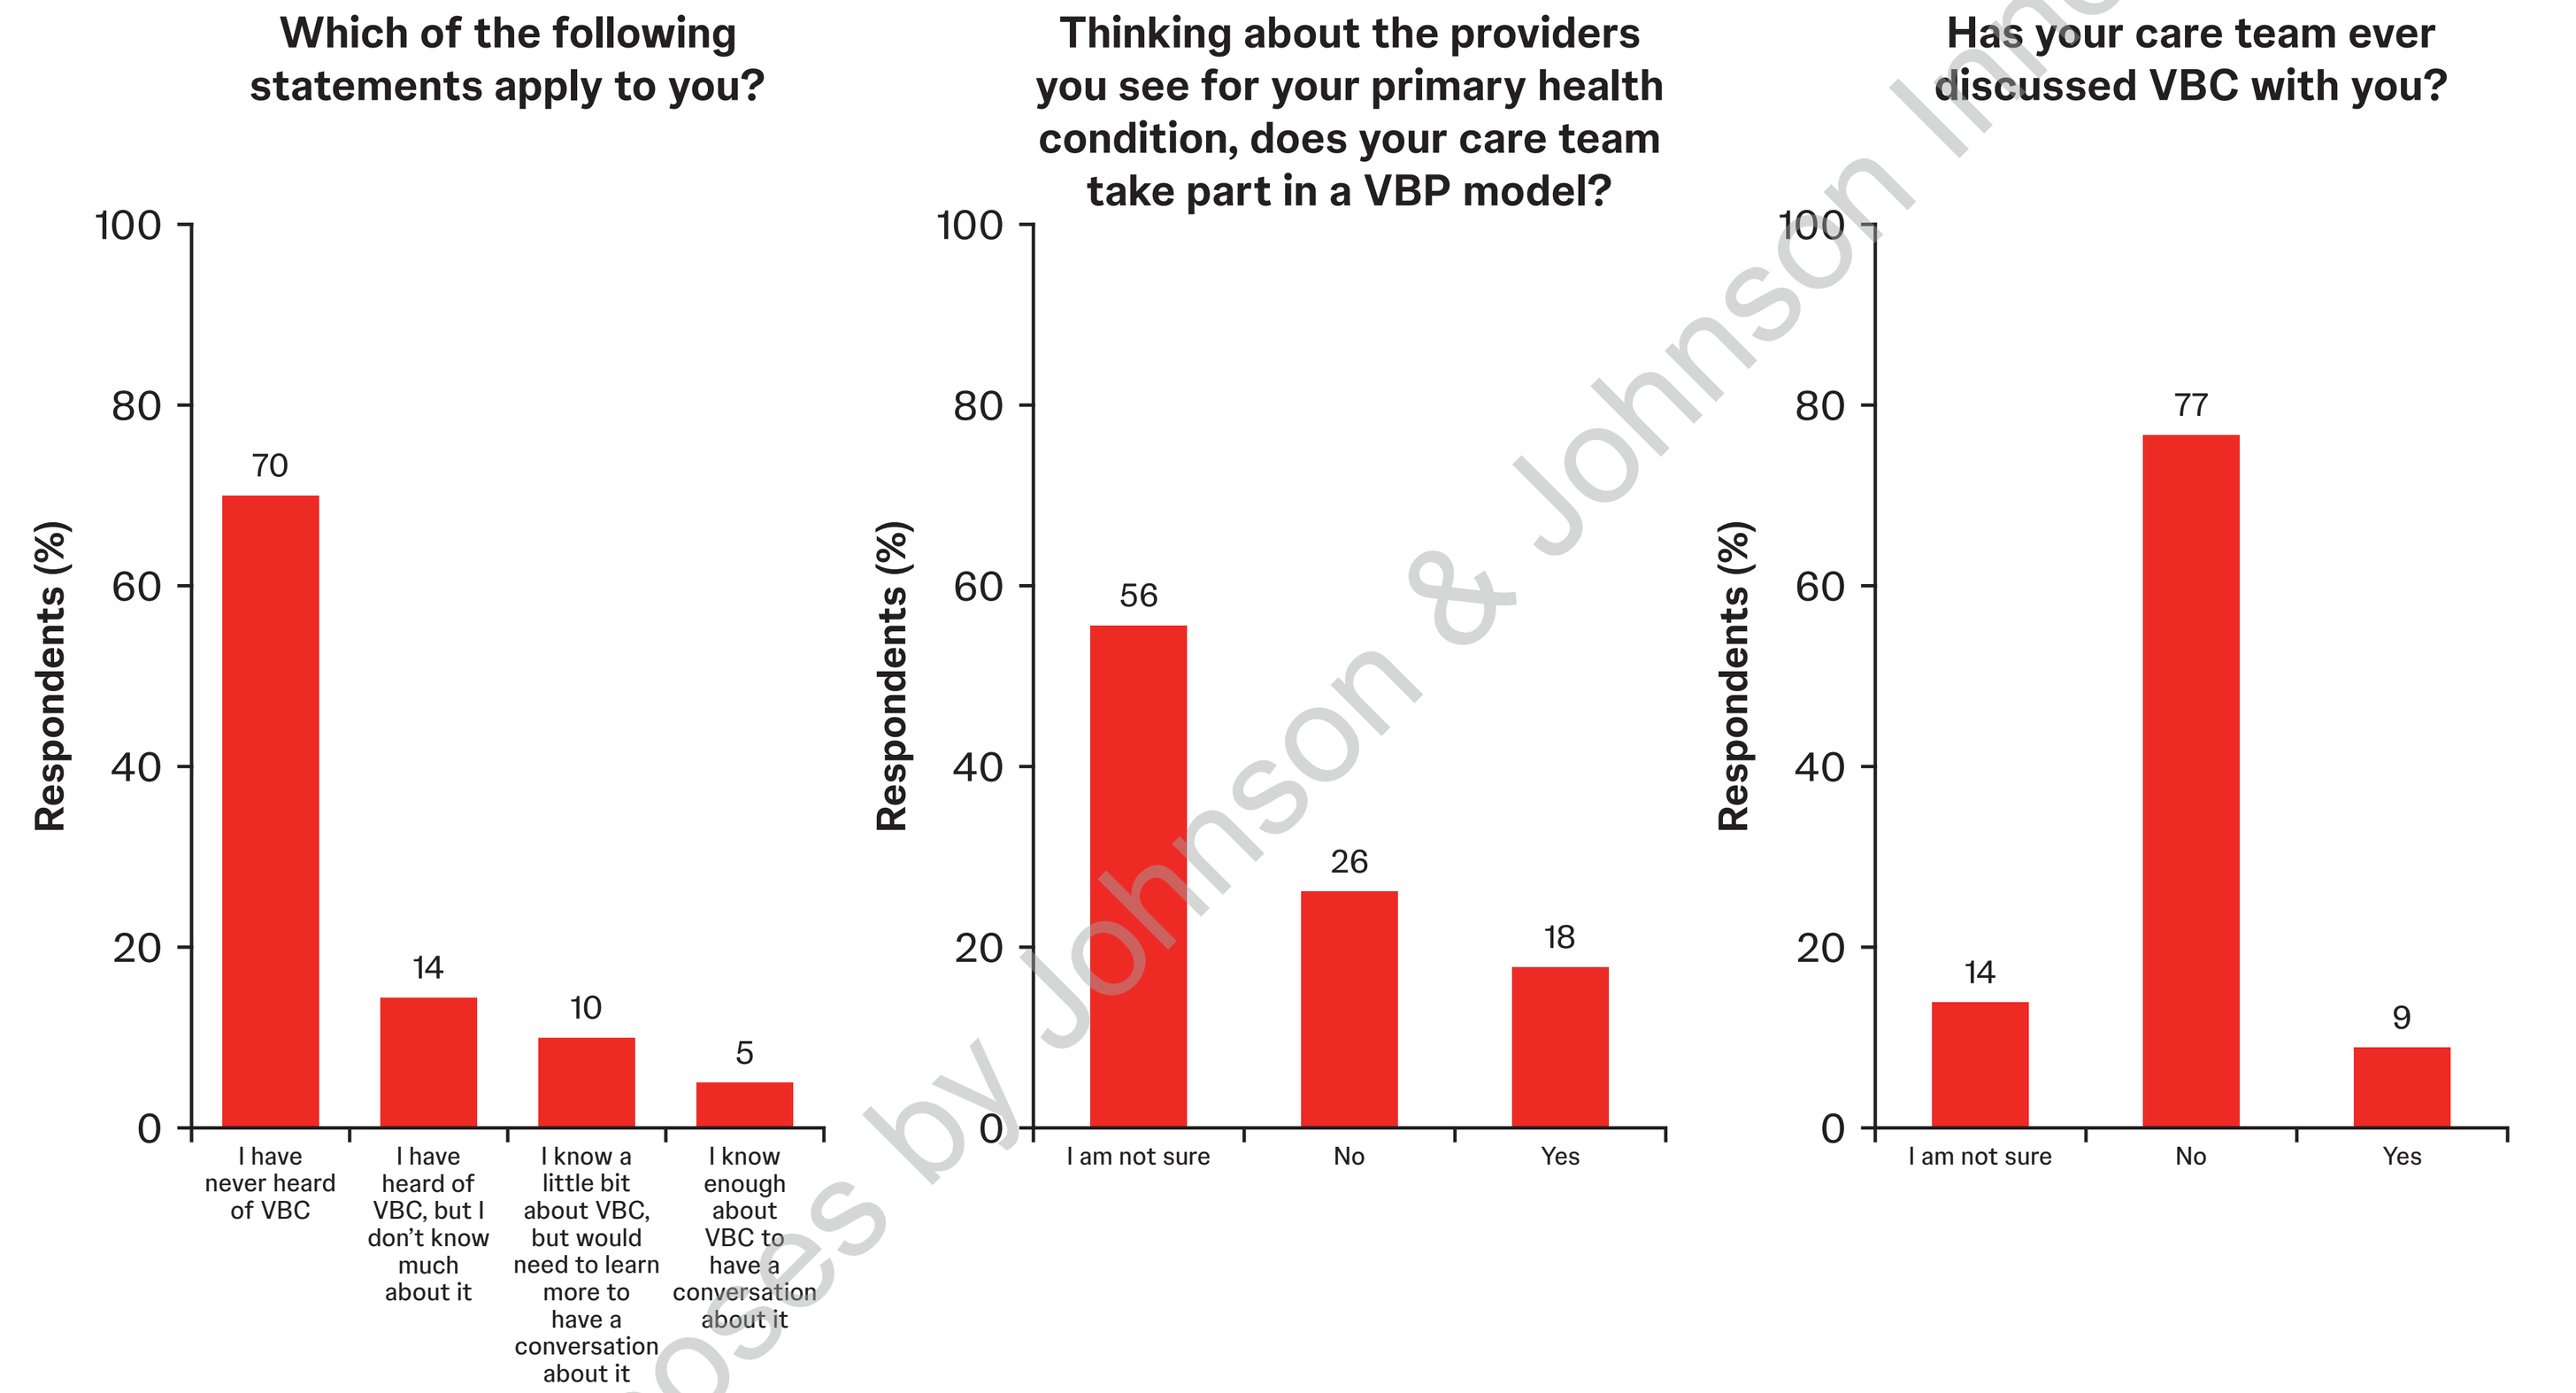
Which of the following statements apply to you?
Thinking about the providers you see for your primary health condition, does your care team take part in a VBP model?
Has your care team ever discussed VBC with you?
Respondents (%)
0
20
40
60
80
100
70
14
10
5
I have never heard of VBC
I have heard of VBC, but I don't know much about it
I know a little bit about VBC, but would need to learn more to have a conversation about it
I know enough about VBC to have a conversation about it
56
26
18
I am not sure
No
Yes
14
77
9

Percentages are rounded and may not add up to 100. **VBC**, value-based care; **VBP**, value-based payment.

TABLE 2: Themes relating to perceptions of VBC

Opinion	Themes	Illustrative quote
Concerned about VBC (48% of respondents)	• Costs will be prioritized over patient care • Distrust of insurance companies • Harm to personalization of patient care • Reduction in patient autonomy • Interference in provider–patient relationships • Reduced quality of care	*"I don't want anyone who is deciding on my quality of life being inspired to make decisions based on costs to an insurance company."*
Unsure about VBC (44% of respondents)	• Not knowing enough to have an opinion • Feeling uncertain about how VBC works	*"I lack enough knowledge to know how it could affect my care."*
Excited about VBC (8% of respondents)	• Potential to address affordability issues in healthcare • Support for individualized care and holistic treatment as a means to improve quality • Interest in trying a new approach	*"Efficient, coordinated care can reduce unnecessary or avoidable services and prevent costly complications, potentially lowering the overall cost of healthcare."*

VBC, value-based care.

- The most common metrics of quality prioritized by respondents were efficacy and safety/tolerability; however, mental health resources and use of electronic health records and electronic patient-reported outcomes—metrics of quality typically used in oncology VBP models—were considered less important by respondents (**Figure 2**)
- Additionally, when respondents were asked to rank six considerations for cost containment in treatment recommendations, efficacy was again the top priority (89% ranking top three); costs to patients ranked fifth (33%) and costs to insurance companies ranked sixth (9%) (**Figure 3**)

FIGURE 2: Perceptions of quality in care (n=830)

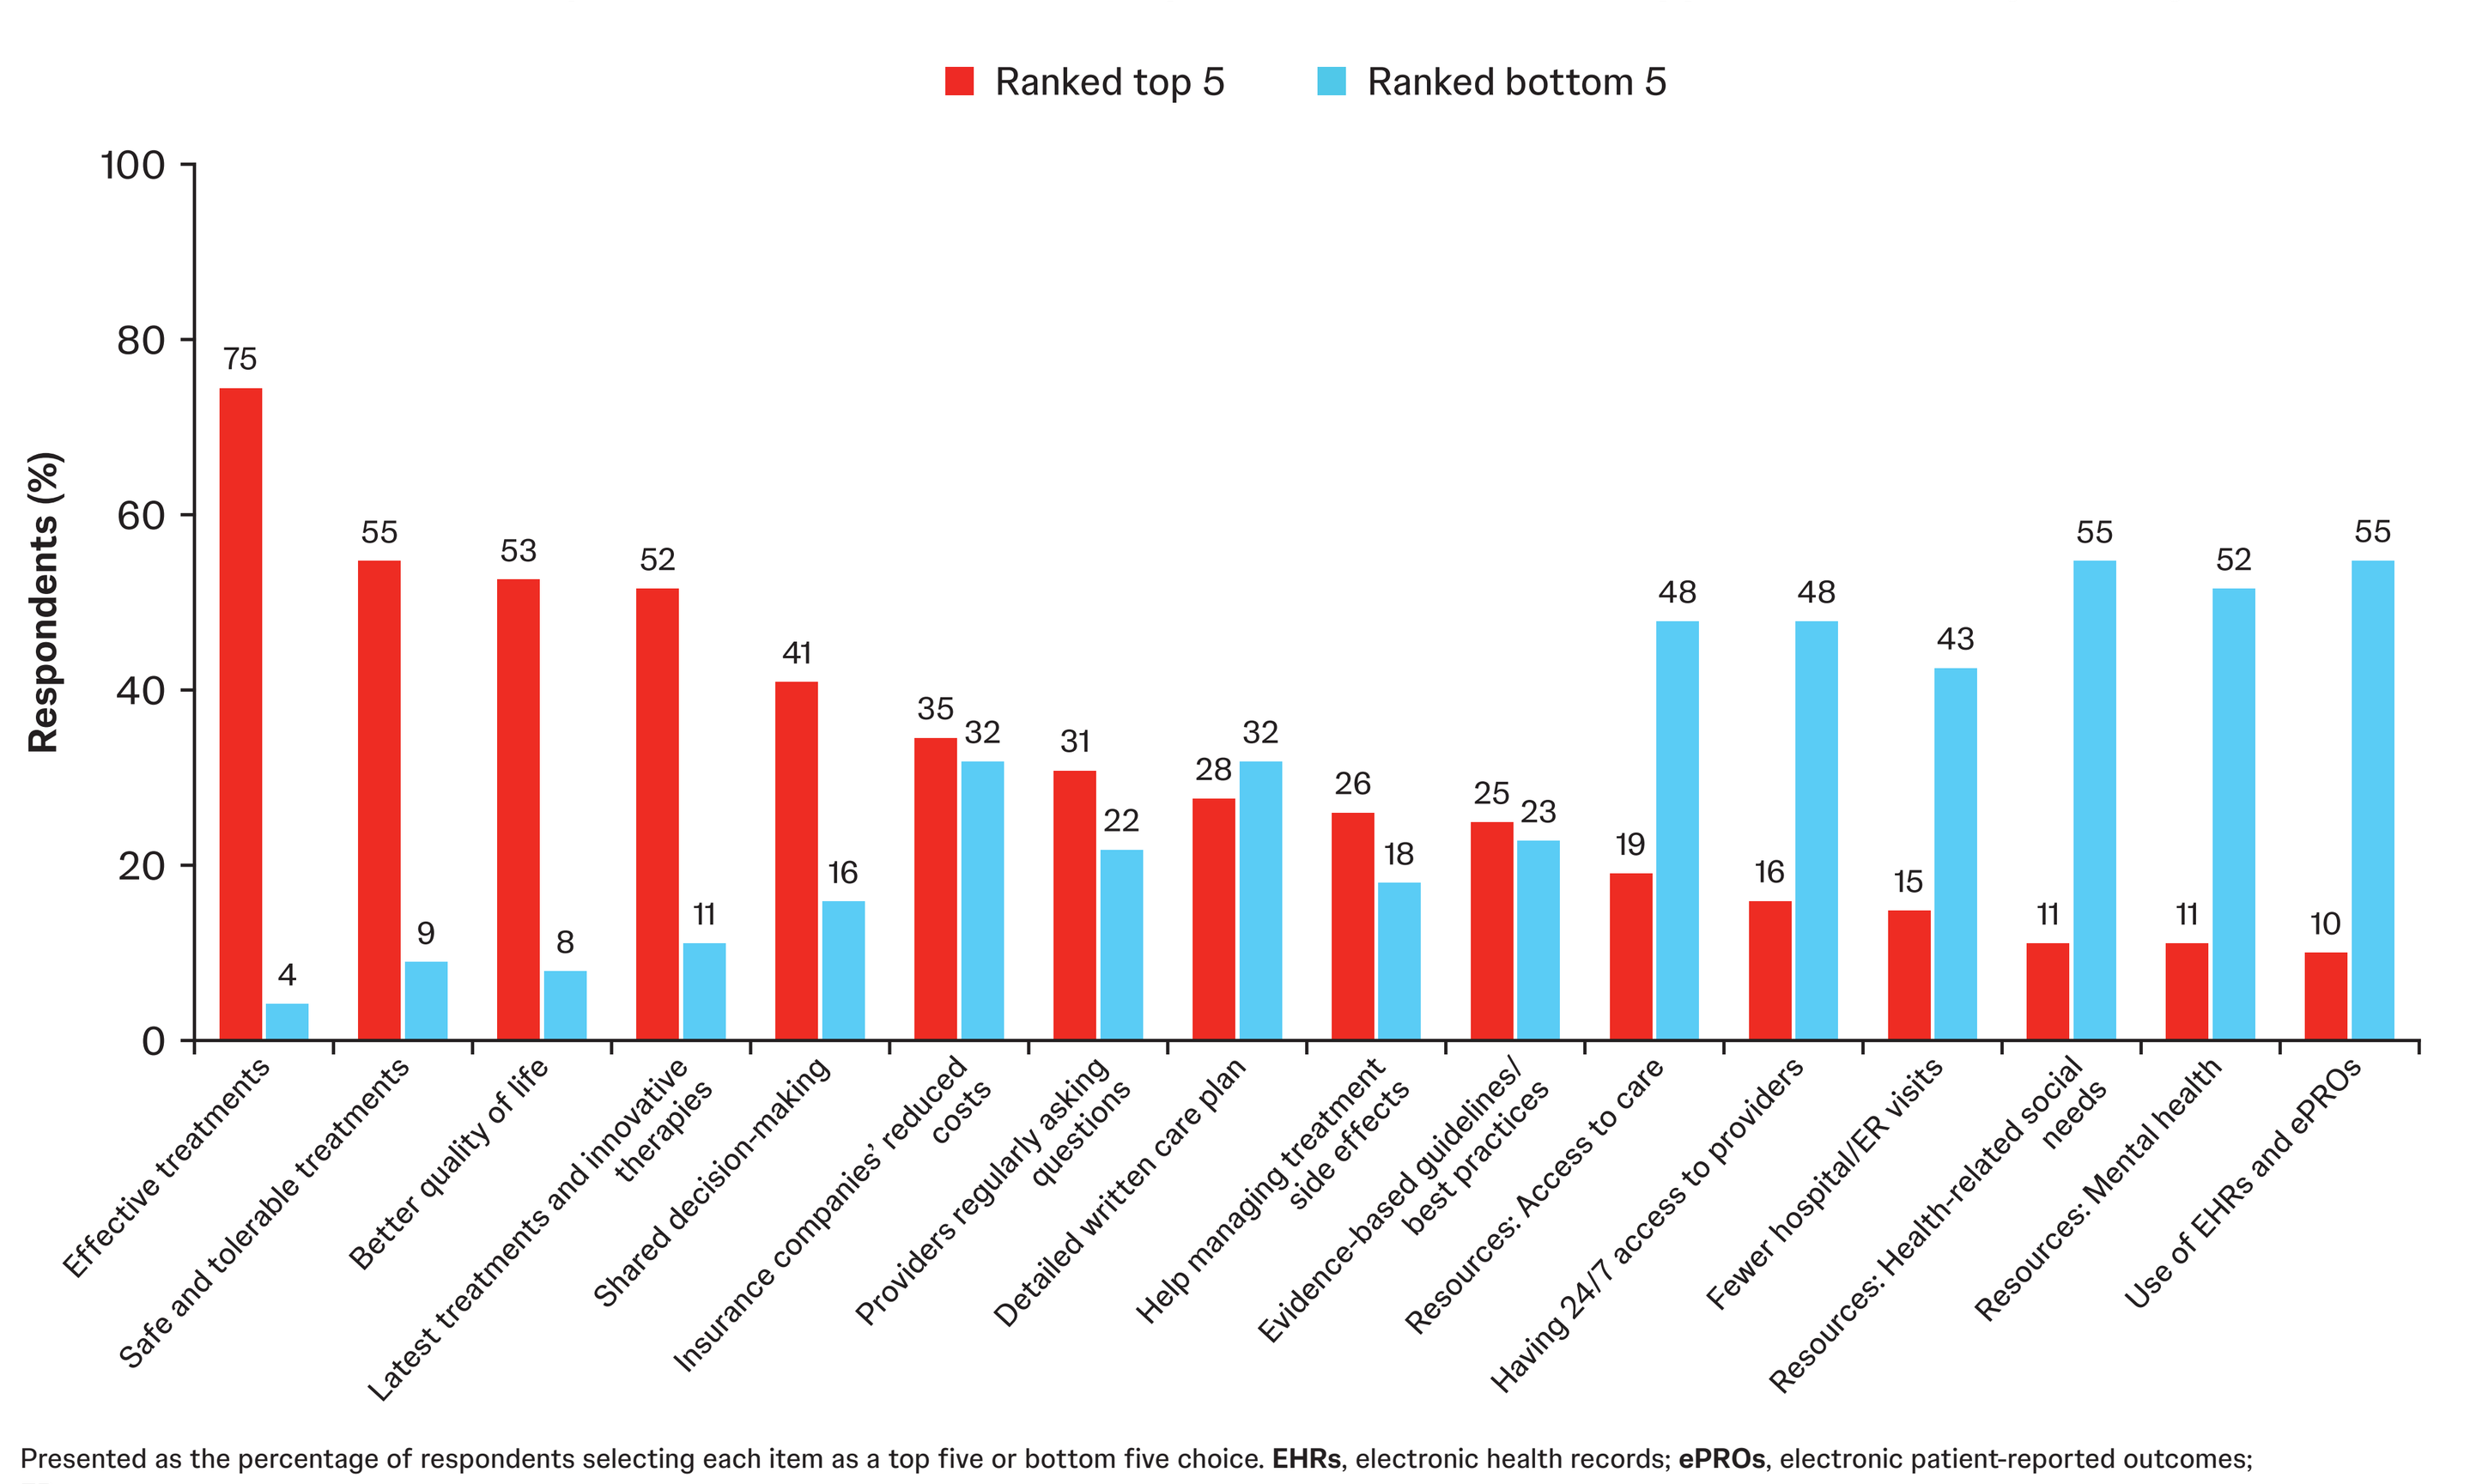
Which are the top five and bottom five most important outcomes and support services to you and your care?
Ranked top 5
Ranked bottom 5
Respondents (%)
0
20
40
60
80
100
75
4
55
9
53
8
52
11
41
16
35
32
31
22
28
32
26
18
25
23
19
48
16
48
15
43
11
55
11
52
10
55
Effective treatments
Safe and tolerable treatments
Better quality of life
Latest treatments and innovative therapies
Shared decision-making
Insurance companies' reduced costs
Providers regularly asking questions
Detailed written care plan
Help managing treatment side effects
Evidence-based guidelines/ best practices
Resources: Access to care
Having 24/7 access to providers
Fewer hospital/ER visits
Resources: Health-related social needs
Resources: Mental health
Use of EHRs and ePROs

Presented as the percentage of respondents selecting each item as a top five or bottom five choice. **EHRs**, electronic health records; **ePROs**, electronic patient-reported outcomes; **ER**, emergency room.

FIGURE 3: Perceptions of cost containment (n=830)

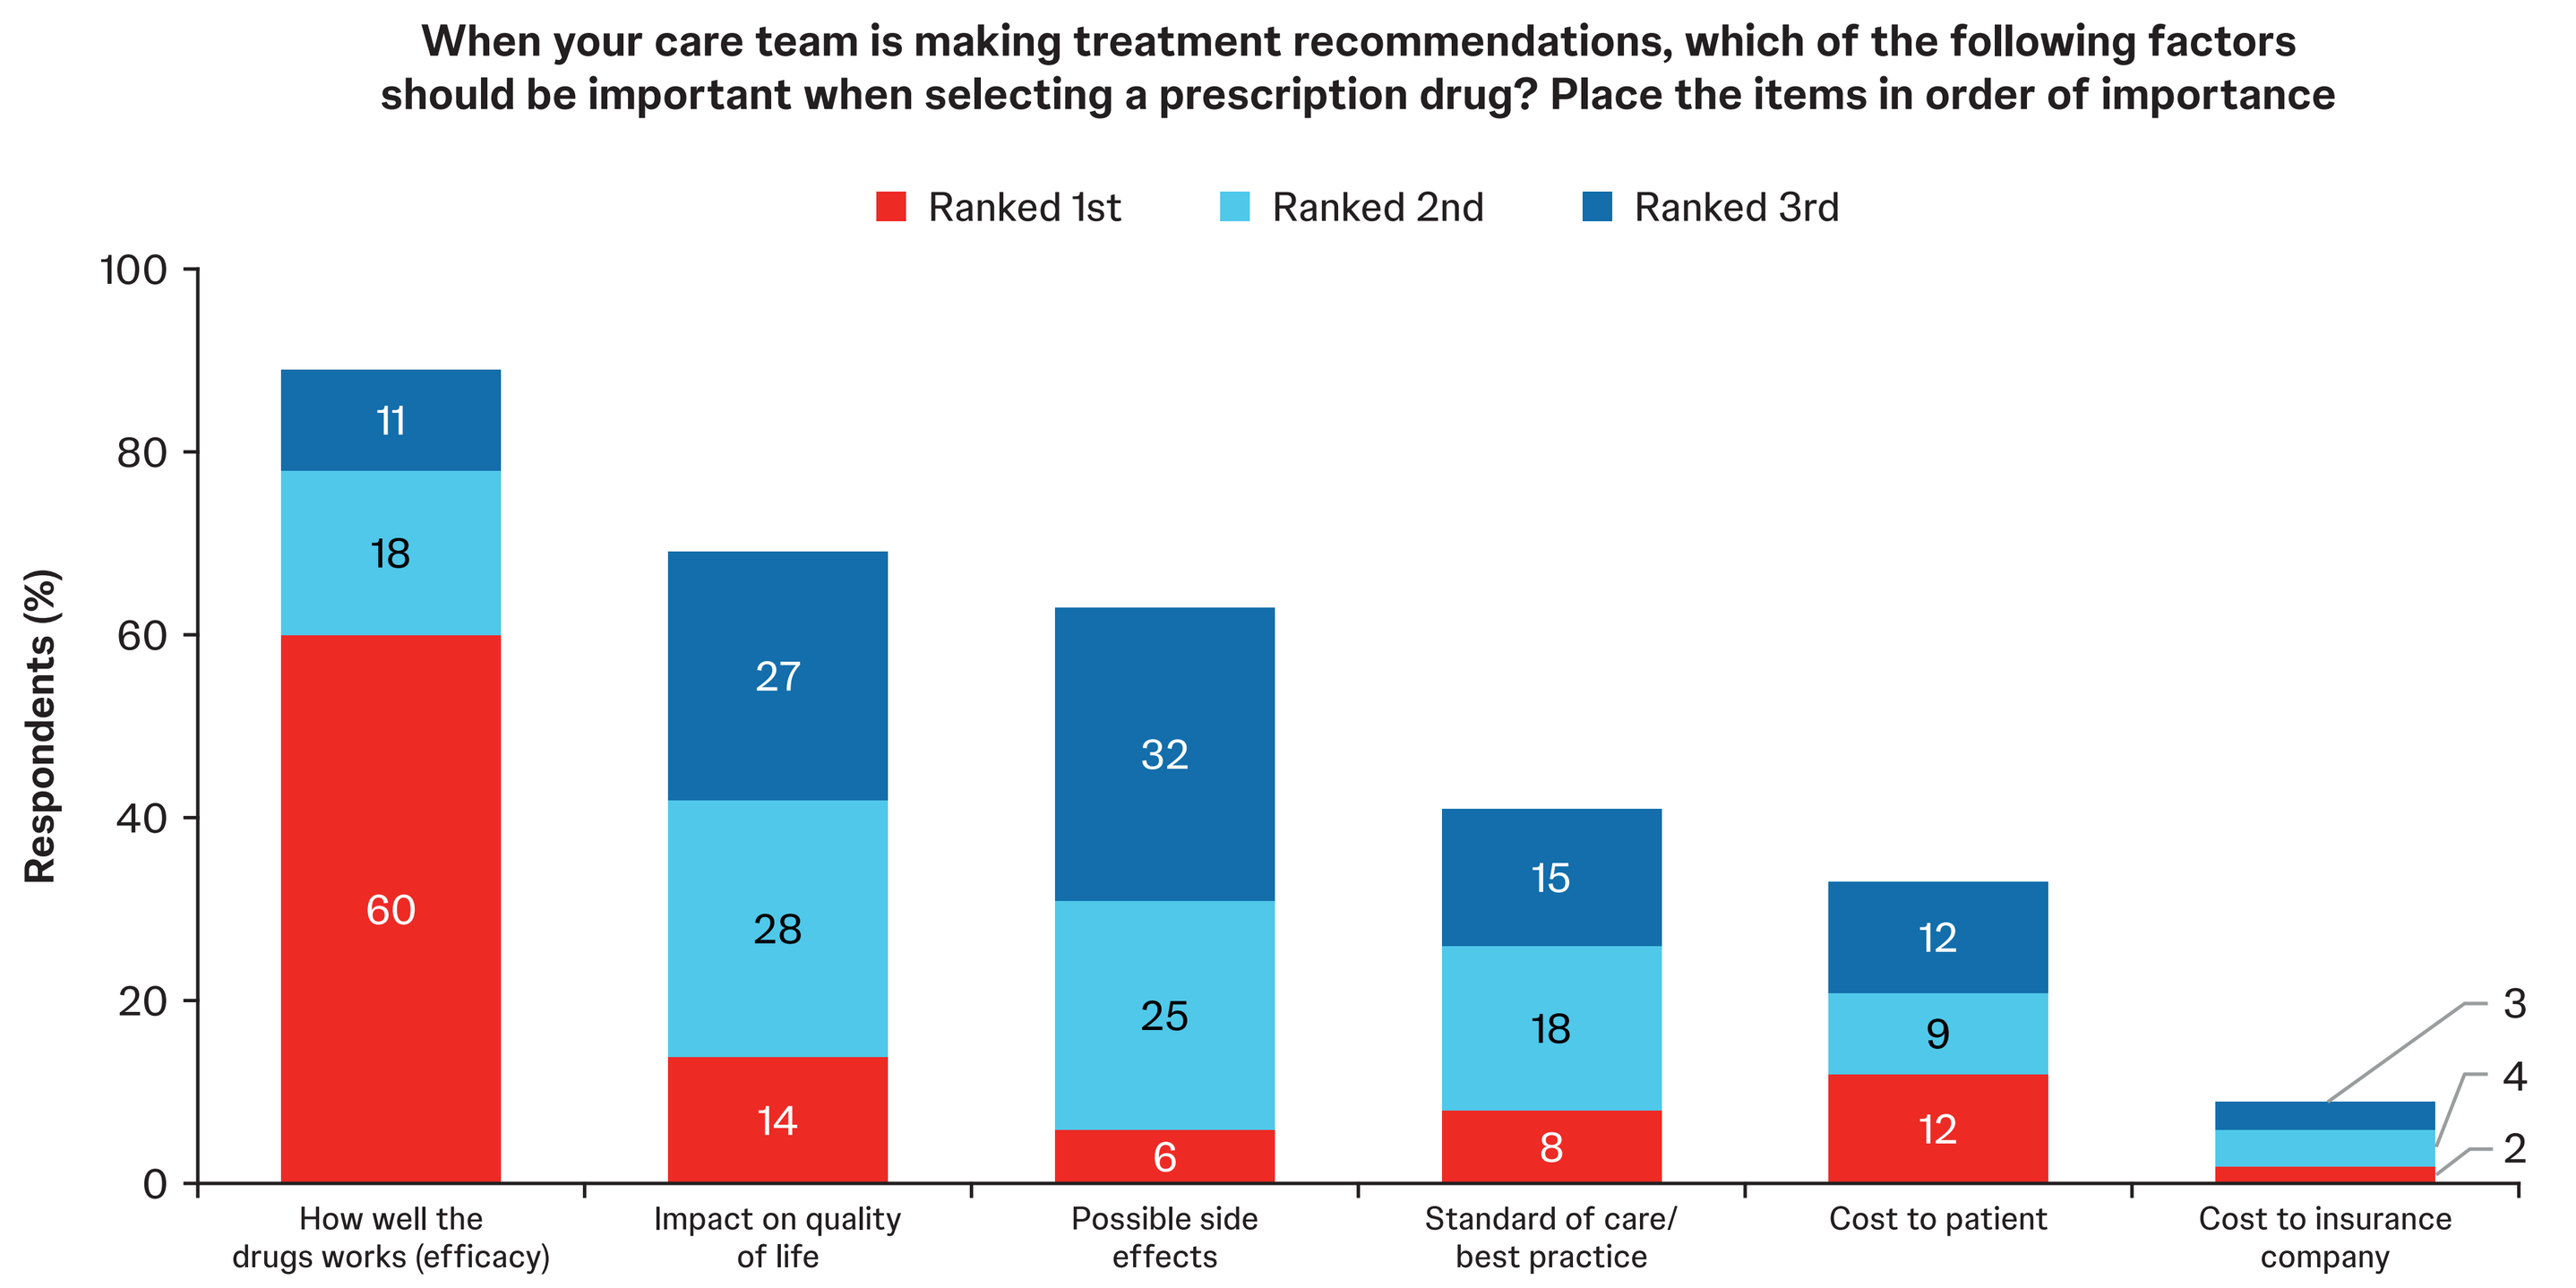
When your care team is making treatment recommendations, which of the following factors should be important when selecting a prescription drug? Place the items in order of importance
Ranked 1st
Ranked 2nd
Ranked 3rd
Respondents (%)
0
20
40
60
80
100
60
18
11
14
28
27
6
25
32
8
18
15
12
9
12
2
4
3
How well the drugs works (efficacy)
Impact on quality of life
Possible side effects
Standard of care/ best practice
Cost to patient
Cost to insurance company

Presented as the percentage of respondents selecting each item as a top three choice.

C22

Key takeaway

Current quality metrics in national VBP models are misaligned with the priorities and perspectives of financially underserved patients

Conclusions

- **Awareness of and familiarity with VBC were low among the 830 survey respondents with cancer from the United States**
- **The respondents wanted providers to prioritize efficacy and safety/tolerability when making treatment recommendations**
- **The preferences and priorities of patients living with cancer are a critical consideration for the meaningful design and delivery of patient-centered VBP models in oncology**

Acknowledgments

This study was funded by Johnson & Johnson. Medical writing support was provided by Adeline Rosenberg, MSc, on behalf of Twist Medical, and was funded by Johnson & Johnson.

Disclosures

RSMA, **KDG**, and **AB** are employees of the Patient Advocate Foundation and have no conflicts to disclose. **KDB** is an employee of and holds stock in Johnson & Johnson, a publicly traded company. **CB** is an employee of Uncommon Insights, LLC, which derives a portion of its profits from interactions with pharmaceutical sponsors.

Oncology

Scan the QR code

Copies of this poster obtained through Quick Response (QR) Code are for personal use only and may not be reproduced without permission from ASCO or the author of this poster.

https://www.asco.org/abstracts-presentations/268436

Presented at the American Society of Clinical Oncology (ASCO) Quality Care Symposium; Boston, MA, USA; October 16–17, 2026

REFERENCES:

1. Balch A, et al. *Health Aff Sch.* 2026;4(3):qxag040. **2.** Patel TA, et al. *J Natl Cancer Inst.* 2023;115:125–30. **3.** Centers for Medicare & Medicaid Services. EOM (Enhancing Oncology Model). https://www.cms.gov/priorities/innovation/innovation-models/eom. Accessed August 12, 2026.

This material is distributed for scientific purposes by Johnson & Johnson Innovative Medicine and is not for promotional use.