Evaluation of Concomitant Medications in Advanced Prostate Cancer Patients Receiving Apalutamide: TITAN and SPARTAN Post-Hoc Analysis

Neeraj Agarwal,[1] Geoffrey Gotto,[2] Alvaro Juarez Soto,[3] Nishant Sangole,[4] Amitabha Bhaumik,[5] Silva Libohova,[4] Alex Yu,[5] Kim Chi[6]

[1]University of Utah Health Hospitals and Clinics, Salt Lake City, UT, USA; [2]University of Calgary, Calgary, AB, Canada; [3]Hospital Universitario de Jerez de la Frontera, Cádiz, Spain; [4]Johnson & Johnson, Raritan, NJ, USA; [5]Johnson & Johnson, Titusville, NJ, USA; [6]BC Cancer and Vancouver Prostate Centre, Vancouver, BC, Canada

Key Takeaway

Apalutamide and other ARPIs are known to have DDIs. In two post-hoc analyses of >2,200 patients with advanced prostate cancer from the TITAN and SPARTAN trials, commonly used concomitant medications, when combined with APA, did not demonstrate clinically meaningful impact on TEAE burden related to individual classes of concomitant medications under standard clinical monitoring

Conclusions

In separate exploratory post-hoc analyses of TITAN and SPARTAN, concomitant use of commonly prescribed medications was frequent and reflected polypharmacy typical of real-world practice

The incidence rates of TEAEs influenced by DDIs associated with 8 common classes of concomitant medications, when co-administered with APA, were comparable with those in patients who did not receive APA

Please scan QR code

Poster

©2026 American Society of Clinical Oncology, Inc. Reused with permission.
This poster was accepted and previously presented at the 2026 ASCO Annual Meeting. All rights reserved.

Copies of this presentation obtained through Quick Response (QR) Codes are for personal use only and may not be reproduced without permission from ASCO® or the author of this presentation.

Acknowledgments

Writing assistance was provided by Tiffany Brake, PhD, funded by Johnson & Johnson. Editorial support was provided by Jennifer Han, MS, of Johnson & Johnson. Layout design was provided by Samita Warang, PhD, of SIRO Medical Writing Ltd., funded by Johnson & Johnson. These studies and the post-hoc analysis are sponsored by Johnson & Johnson.

Disclosures

NA has received consultancy fees from Astellas, AstraZeneca, Aveo, Bayer, Bristol Myers Squibb, Calithera, Clovis, Eisai, Eli Lilly, EMD Serono, Exelixis, Foundation Medicine, Genentech, Gilead, Janssen, Merck, MEI Pharma, Nektar, Novartis, Pfizer, Pharmacyclics, and Seattle Genetics; research funding (payment to institution) from Astellas, AstraZeneca, Bavarian Nordic, Bayer, Bristol Myers Squibb, Calithera, Celldex, Clovis, Eisai, Eli Lilly, EMD Serono, Exelixis, Genentech, Gilead, Glaxo Smith Kline, Immunomedics, Janssen, Medivation, Merck, Nektar, New Link Genetics, Novartis, Pfizer, Prometheus, Rexahn, Roche, Sanofi, Seattle Genetics, Takeda, and Tracon. **GG** reports consulting/advisory roles with Astellas Pharma, AstraZeneca, Bayer, Bristol Myers Squibb, EMD Serono, Ferring, Janssen, Merck, Novartis, Pfizer and Tolmar; honoraria from Astellas Pharma, AstraZeneca, Bayer, Janssen, Merck and Tolmar; and travel expenses from Janssen and Pfizer. **AS** received consultancy fees from Astellas, Bayer, and Janssen; participated as an investigator for Bayer and Janssen; and received research funding from Janssen. **NS, AB, SL,** and **AY** are full-time employees of Johnson & Johnson and may own company stock. **KNC** reports research grants (payment to institution) from Astellas, AstraZeneca, Janssen, Merck, Novartis, Pfizer, Point Biopharma, Roche, and Sanofi; consulting fees from Astellas, AstraZeneca, Janssen, Merck, Novartis, Pfizer, Point Biopharma, Roche, and Sanofi; payment or honoraria for lectures, presentations, speakers bureaus, manuscript writing or educational events from Astellas, AstraZeneca, Janssen, Merck, Novartis, Pfizer, Point Biopharma, Roche, and Sanofi; payment for expert testimony from AstraZeneca, Janssen, Merck, and Novartis.

Introduction

- Prostate cancer is most common in older men (average age at diagnosis: 66 years),[1] most of whom have at least one comorbidity for which they are also receiving pharmacologic treatment[2-4]
- Apalutamide (APA) is an androgen receptor pathway inhibitor (ARPI) approved for use with androgen-deprivation therapy (ADT) in patients with metastatic castration-sensitive prostate cancer (mCSPC) and nonmetastatic castration-resistant prostate cancer (nmCRPC) based on results from the global, phase 3 TITAN[5,6] and SPARTAN trials[7,8]
- APA is a strong inducer of CYP3A4 and CYP2C19, a weak inducer of CYP2C9, and an inducer of P-glycoprotein, breast cancer resistance protein, and organic anion transporting polypeptide 1B1[9]
- Interactions with APA may alter the pharmacokinetics of commonly used concomitant medications, raising the possibility for treatment-emergent adverse events (TEAEs) due to drug-drug interactions (DDIs)

Objective

- To evaluate whether DDIs with APA impact safety and tolerability by comparing the safety profiles of APA + ADT versus placebo (PBO) + ADT in patients receiving concomitant medications in the TITAN and SPARTAN trials

Methods

- In TITAN (NCT02489318), 1052 patients with mCSPC were randomized 1:1 to receive APA + ADT (N=525) or PBO + ADT (N=527). APA + ADT resulted in significantly longer overall and radiographic progression-free survival versus PBO + ADT, with similar safety profiles and preserved health-related quality of life[5]
- In SPARTAN (NCT01946204), 1207 patients with nmCRPC receiving ongoing ADT were randomized 2:1 to APA (N=806) or matched PBO (N=401). APA + ADT significantly decreased the risk of distant metastasis or death, improved overall survival, and was well tolerated compared with PBO + ADT[8]
- These post-hoc analyses from TITAN and SPARTAN examined the occurrence of TEAEs that may be influenced by DDIs when common concomitant medication classes are co-administered with APA. The incidence rates of these TEAEs were compared descriptively between the treatment arms
- The selected drug classes included some of the most frequently prescribed agents that have the potential for DDIs with APA
 - Our analyses included the 8 most frequently used concomitant medication classes in TITAN and SPARTAN: anticoagulants, antidiabetics, antihypertensives, corticosteroids, gabapentinoids, macrolides, proton pump inhibitors, and statins
- Because the median treatment duration in the APA + ADT arm exceeded that of the PBO + ADT arm by 12 to 26 months across the TITAN and SPARTAN trials, we analyzed exposure-adjusted incidence rate of TEAEs per 100 patient-years (PY) to account for the imbalance in time at risk

Results

- Patients receiving 1 or more of the 8 most commonly used concomitant medication classes had baseline characteristics generally consistent with the overall TITAN and SPARTAN safety populations (**Supplementary Table 1**)
- Exposure-adjusted rates of any-grade and grade 3/4 TEAEs were comparable between with APA + ADT and PBO + ADT among patients receiving commonly used concomitant medications in both TITAN (**Figure 1A**) and SPARTAN (**Figure 1B**), with no signal of increased TEAE burden associated with DDIs

Figure 1. Incidence rates of TEAEs influenced by DDIs[a] associated with concomitant medication use in patients receiving APA versus those who did not receive APA (after adjusting for treatment exposure) in TITAN (A) and SPARTAN (B)

A.

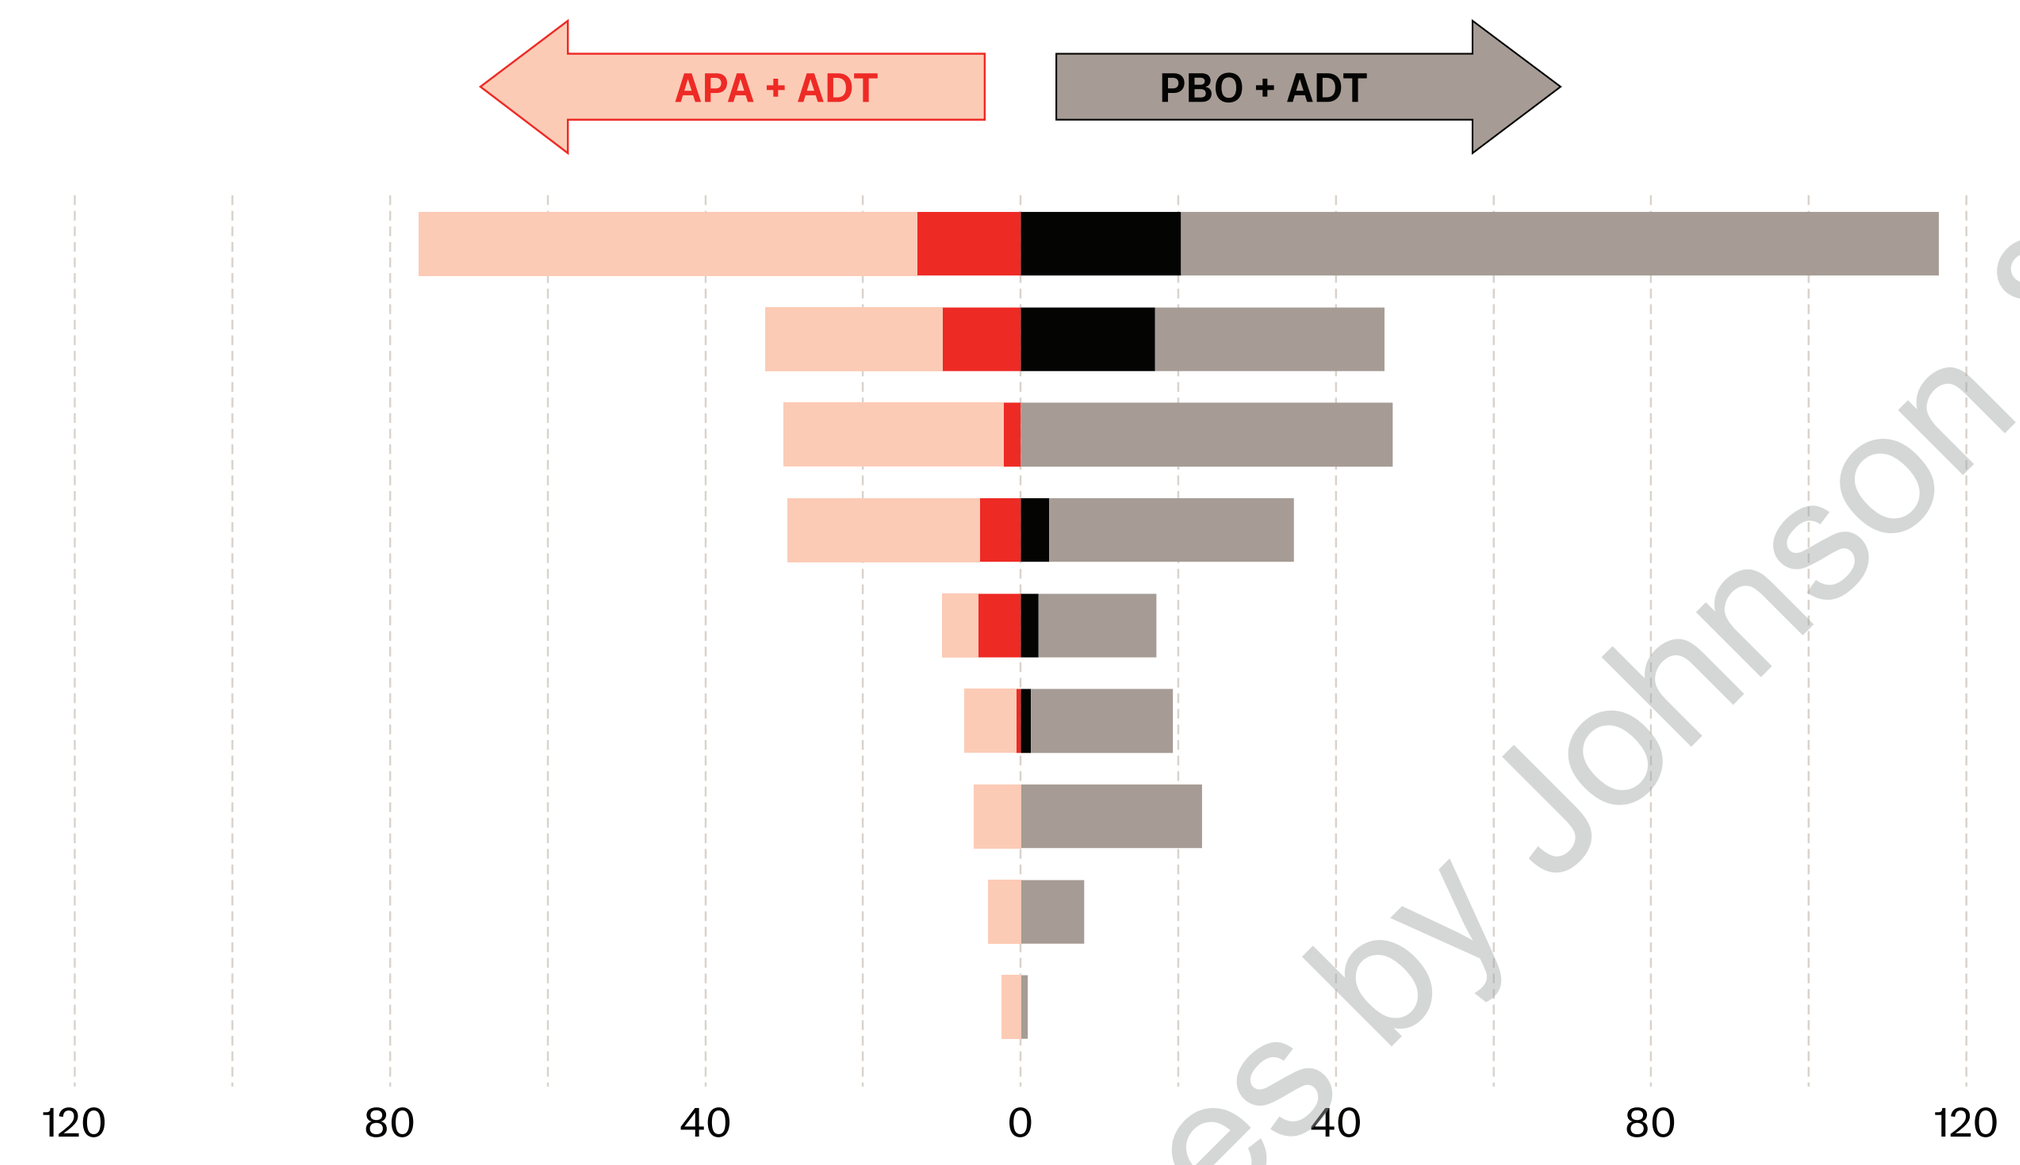
APA + ADT
PBO + ADT
120
80
40
0
40
80
120

	APA + ADT			PBO + ADT		
	n[b]	Any Grade TEAEs/ 100 PY[c]	Grade 3-4 TEAEs/ 100 PY[c]	n[b]	Any Grade TEAEs/ 100 PY[c]	Grade 3-4 TEAEs/ 100 PY[c]
Any concomitant medication[d]	351	76.5	13.2	320	116.5	20.3
Antihypertensive	215	32.5	10.0	181	46.1	17.0
Macrolide	28	30.1	2.2	23	47.2	0
Antidiabetic	64	29.7	5.2	57	34.6	3.6
Anticoagulant	39	10.0	5.4	31	17.2	2.2
Corticosteroid	78	7.2	0.6	59	19.3	1.3
Gabapentinoid[e]	14	6.0	0	21	23.0	0
Statin	125	4.2	0	82	8.0	0
Proton pump inhibitor	92	2.5	0	77	0.9	0

B.

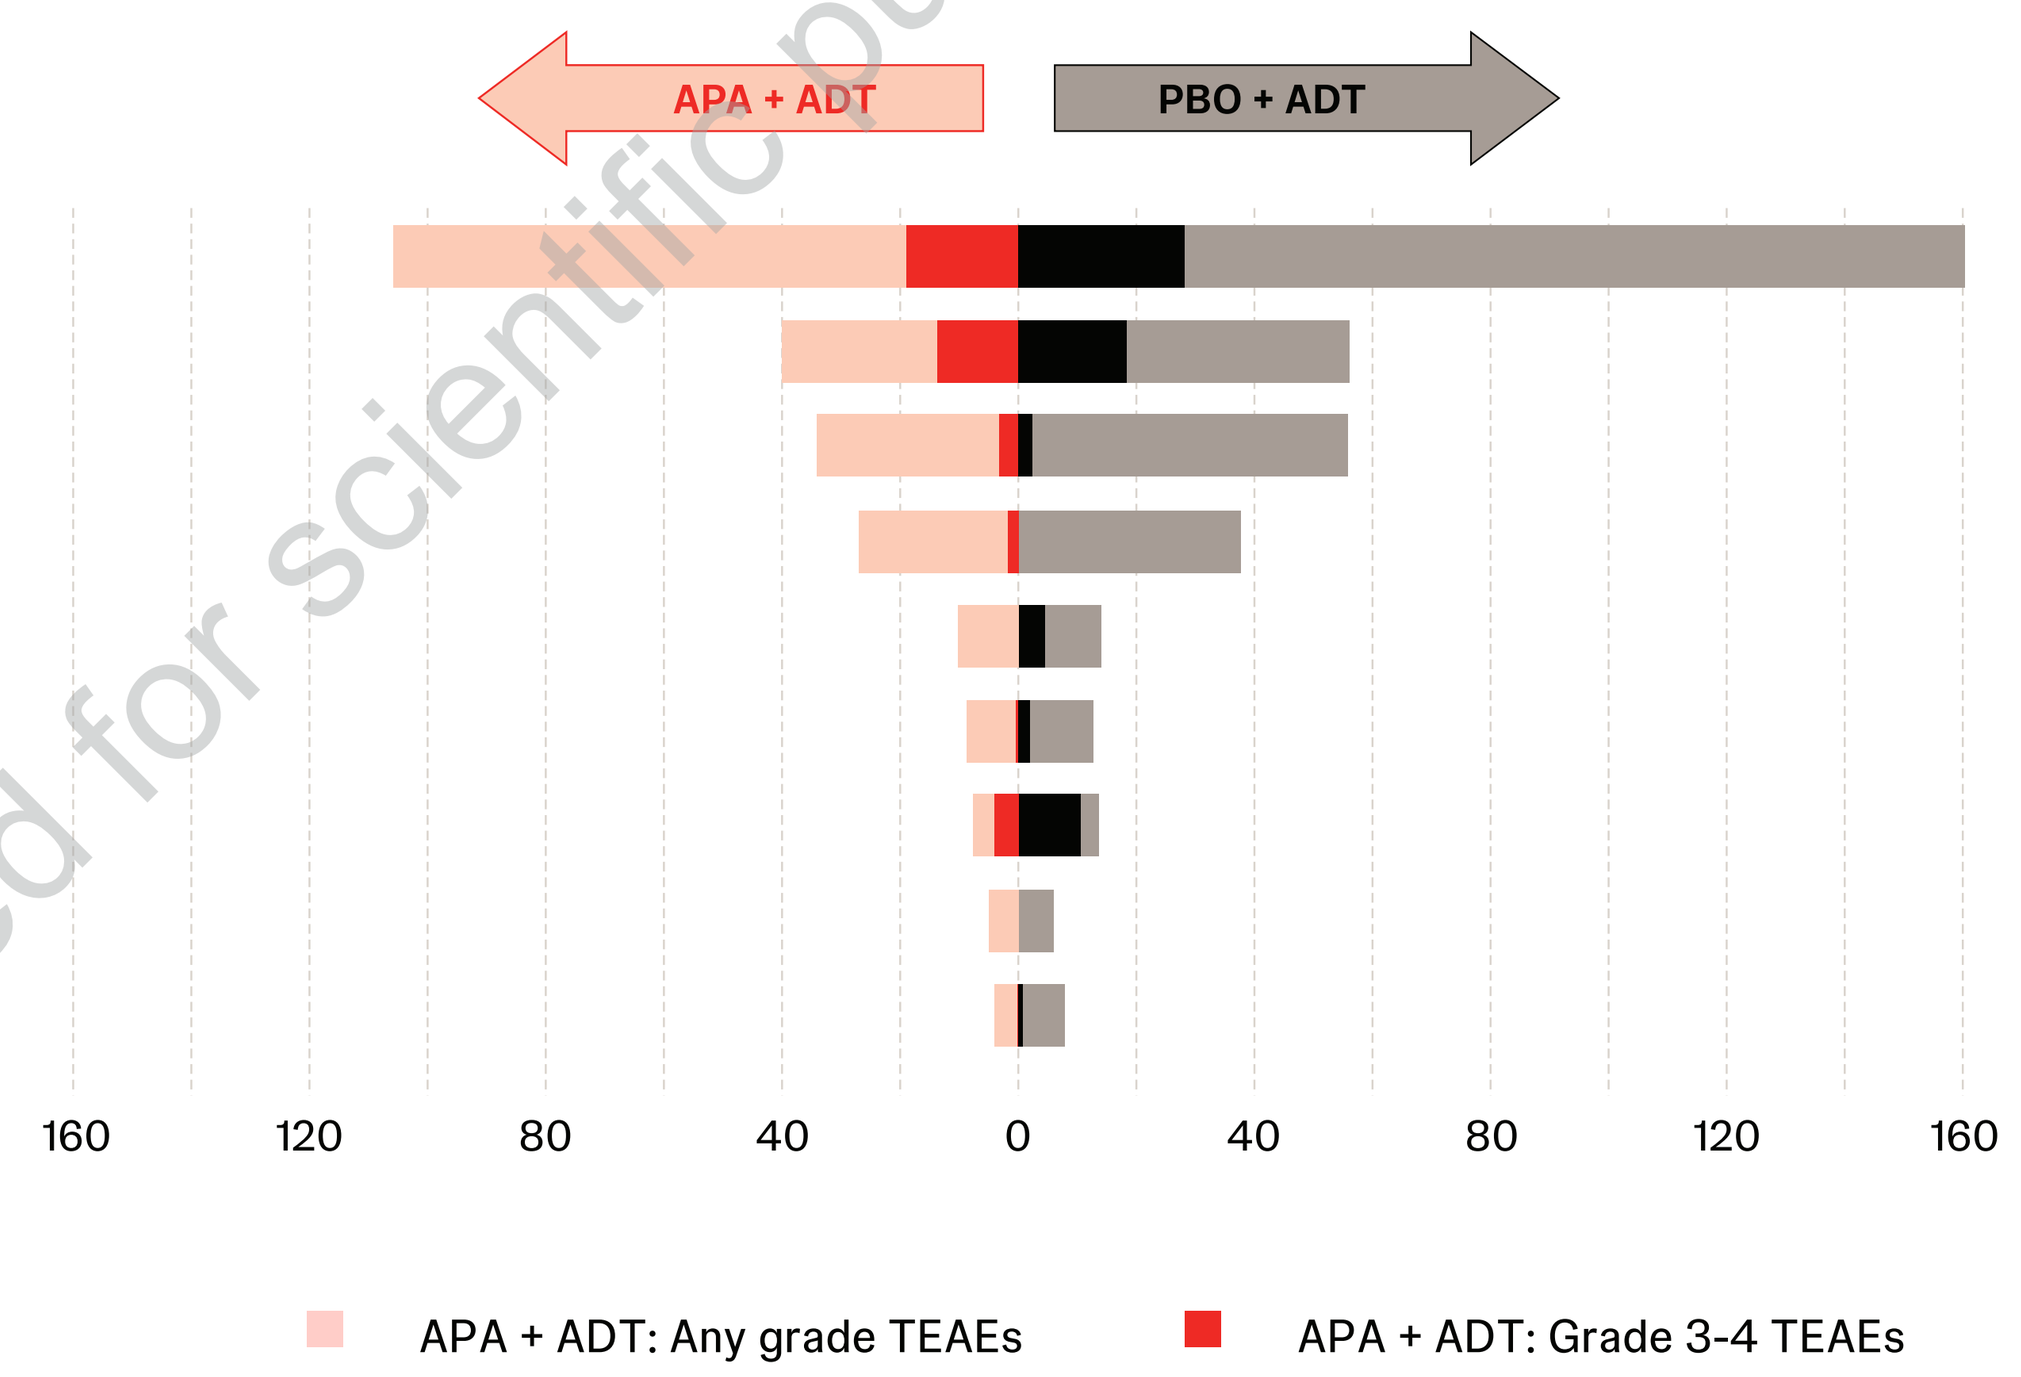
APA + ADT
PBO + ADT
160
120
80
40
0
40
80
120
160
APA + ADT: Any grade TEAEs
APA + ADT: Grade 3-4 TEAEs
PBO + ADT: Any grade TEAEs
PBO + ADT: Grade 3-4 TEAEs

	APA + ADT			PBO + ADT		
	n[b]	Any Grade TEAEs/ 100 PY[c]	Grade 3-4 TEAEs/ 100 PY[c]	n[b]	Any Grade TEAEs/ 100 PY[c]	Grade 3-4 TEAEs/ 100 PY[c]
Any concomitant medication[d]	589	105.8	19.0	273	160.3	28.2
Antihypertensive	344	39.9	13.7	163	56.1	18.5
Antidiabetic	98	34.1	3.3	37	55.7	2.4
Macrolide	54	27.0	1.8	16	37.8	0
Gabapentinoid[e]	31	10.2	0	16	14.1	4.7
Corticosteroid	86	8.7	0.4	33	12.8	2.1
Anticoagulant	109	7.6	4.0	54	13.6	10.6
Statin	272	5.0	0	126	6.0	0
Proton pump inhibitor	209	4.0	0.2	86	7.9	0.9

[a]Although preferred terms chosen for the analyses of TEAEs associated with individual classes of concomitant medications could be influenced by DDIs with APA, they could also be due to class effects of concomitant medication alone. [b]Number of patients receiving concomitant medications for each treatment arm per study. [c]Number of distinct TEAEs per 100 PY of exposure can be >100: calculated as 100×(# of distinct events with that preferred term)/total PY of exposure, with total days of exposure=365.25. [d]Among 8 frequently used medication classes: anticoagulant, antidiabetic, antihypertensive, corticosteroid, gabapentinoid, macrolide, proton pump inhibitor, statin. [e]Gabapentin was administered to patients for indications not related to seizure; patients with seizures were excluded from both trials.
ADT, androgen deprivation therapy; APA, apalutamide; PBO, placebo; PY, patient-year; TEAE, treatment-emergent adverse event.

- Antihypertensive agents were the most frequently used concomitant medication class. Among antihypertensive recipients, exposure-adjusted TEAE rates were comparable between APA + ADT and PBO + ADT in TITAN (**Table 1**); SPARTAN findings are shown in **Supplementary Table 2**

Table 1. Incidence rates of TEAEs influenced by DDIs[a] associated with concomitant antihypertensive[b] use in patients receiving APA versus those who did not receive APA (after adjusting for treatment exposure) in TITAN

	APA + ADT (n=215)		PBO + ADT (n=181)	
	Any grade TEAEs/100 PY[c]	Grade 3/4 TEAEs/100 PY[c]	Any grade TEAEs/100 PY[c]	Grade 3/4 TEAEs/100 PY[c]
Overall TEAEs	32.5	10.0	46.1	17.0
Vascular disorders	17.1	8.4	28.1	14.2
Hypertension	16.6	8.4	26.7	14.2
Hypotension	0.5	0	0.7	0
Orthostatic hypotension	0	0	0.7	0
Metabolism & nutrition disorders	10.4	1.0	12.5	2.1
Hyperkalemia	6.7	0.2	4.2	1.0
Hypokalemia	1.8	0.7	3.5	0.3
Hypercalcemia	0.7	0	0.7	0
Hypocalcemia	0.7	0	0	0
Hyponatremia	0.5	0.2	3.8	0.7
Hypomagnesemia	0	0	0.3	0
Nervous system disorders	4.0	0.7	4.2	0
Dizziness	4.0	0.7	4.2	0
General disorders & administration site conditions	1.0	0	1.4	0.7
Peripheral swelling	1.0	0	1.4	0.7
Cardiac disorders	0	0	0	0
Tachycardia	0	0	0	0
Bradycardia	0	0	0	0

[a]Although preferred terms chosen for the analysis of TEAEs associated with concomitant antihypertensives could be influenced by DDIs with APA, they could also be due to class effects of concomitant antihypertensive alone. [b]Antihypertensive medications examined: angiotensin-converting enzyme inhibitors, angiotensin II receptor blockers, beta blockers, calcium channel blockers, and thiazide diuretics. [c]Exposure-adjusted incidence rates of TEAEs are expressed per 100 patient-years for the treatment arm. Each rate is calculated by dividing the number of distinct TEAEs (overall or within a specified SOC or PT) by total patient-years of exposure (total exposure days divided by 365.25) and multiplying the result by 100.
ADT, androgen deprivation therapy; APA, apalutamide; PBO, placebo; PT, preferred term; PY, patient-year; TEAE, treatment-emergent adverse event.

Limitations

- Subgroup comparisons were descriptive in nature, and the study was not powered for definitive between-subgroup inference
- The concomitant medications co-administered with APA bearing DDI risk and associated TEAEs were selected post hoc
- Although preferred terms chosen for the analyses of TEAEs associated with individual classes of concomitant medications could be influenced by DDIs with APA, they could also be due to class effects of concomitant medication alone
- Some medications included were not continuations from the baseline. In clinical practice, comprehensive medication reconciliation, individualized management of comorbid conditions, and routine clinical monitoring should be maintained when APA is administered to patients with comorbidities
- As TITAN and SPARTAN enrolled selected trial populations, generalizability to routine clinical practice, particularly patients with more extensive comorbidity and polypharmacy, may be limited

References

1. Spratt DE, et al. *Prostate Cancer Prostatic Dis* 2021;24:647-661. **2.** Benzo RM, et al. *Support Care Cancer* 2023;31:496. **3.** Eaglehouse YL, et al. *Cancer Control* 2026;33:10732748261420506. **4.** Lees J and Chan A. *Lancet Oncol* 2011;12:1249-1257. **5.** Chi KN, et al. *N Engl J Med* 2019;381:13-24. **6.** Chi KN, et al. *J Clin Oncol* 2021;39:2294-2303. **7.** Smith MR, et al. *Eur Urol* 2021;79:150-158. **8.** Smith MR, et al. *N Engl J Med* 2018;378:2542. **9.** ERLEADA® [prescribing information]. Horsham, PA: Janssen Products, LP; 2026. https://www.jnjlabels.com/package-insert/product-monograph/prescribing-information/ERLEADA-pi.pdf.

Prostate Cancer

Originally presented at 2026 ASCO Annual Meeting, Chicago, IL, USA; Encore presentation at JADPRO Live 2026 October 15–18, 2026; Minneapolis, MN, USA

This material is distributed for scientific purposes by Johnson & Johnson Innovative Medicine and is not for promotional use.