5085

Cognition Varies Between Apalutamide + Androgen Deprivation Treatment Responders and Nonresponders

Arun Azad,[1] Marco Antonio Badillo Santoyo,[2] Alicia K. Morgans,[3] Dana E. Rathkopf,[4] Karie Runcie,[5] Geoffrey Gotto,[6] Axel S. Merseburger,[7] Alex Dos Santos,[8*] Jingwei Li,[9] Sukie Shopeju,[8] Meenakshi Chatterjee,[10] Rana Zia Ur Rehman,[11] Daniel Sanchez,[12] Bhavini Patel,[11] Amitabha Bhaumik,[13] Vasanth Thirugnanam,[14] Yanran Huo,[10] Ashita S. Batavia,[8] Mark Wildgust,[8] Neeraj Agarwal[15]

[1]Peter MacCallum Cancer Centre, Melbourne, Australia; [2]Hospital Aranda de la Parra, Guanajuato, Mexico; [3]Dana-Farber Cancer Institute, Boston, MA, USA; [4]Memorial Sloan Kettering Cancer Center, NY, USA; [5]New York-Presbyterian/Columbia University Medical Center, NY, USA; [6]Southern Alberta Institute of Urology, University of Calgary, Calgary, AB, Canada; [7]Universitaetsklinikum Schleswig-Holstein - Campus Luebeck, Germany; [8]Johnson & Johnson, Raritan, NJ, USA; [9]Johnson & Johnson, Neuss, Germany; [10]Johnson & Johnson, Cambridge, MA, USA; [11]Johnson & Johnson, High Wycombe, UK; [12]Johnson & Johnson, Jacksonville, FL, USA; [13]Johnson & Johnson, Titusville, NJ, USA; [14]Johnson & Johnson, Brisbane, Australia; [15]Huntsman Cancer Institute at the University of Utah, Salt Lake City, UT, USA

*Presenting author

Key Takeaway

Cognitive differences between PSA responders and nonresponders are present at baseline and persist during early treatment with apalutamide + ADT in pts with mCSPC

Conclusions

PSA responders demonstrated better visual memory and executive function at baseline and throughout initial treatment with apalutamide + ADT in pts, maintaining a cognitive advantage compared with nonresponders

Significant longitudinal changes in cognitive performance were observed among PSA responders to apalutamide + ADT in pts with mCSPC

Although this retrospective analysis did not assess cognition as a predictive biomarker, these findings suggest that early cognitive performance may support treatment stratification for future prospective studies in mCSPC

Please scan QR code

Poster

Supplementary material

https://www.congresshub.com/Oncology/AM2026/Apalutamide/Santos

Copies of this presentation obtained through Quick Response (QR) Codes are for personal use only and may not be reproduced without permission from ASCO® or the author of this presentation.

Acknowledgments

Writing assistance was provided by Jennifer Venzie, PhD, CMPP (SystemOne), and was funded by Johnson & Johnson. Editorial support was provided by Jennifer Han, MS, CMPP (Johnson & Johnson). Layout design was provided by Sandeep Chavan of SIRO Medical Writing Pvt. Ltd. India, funded by Johnson & Johnson. This study is sponsored by Johnson & Johnson.

Disclosures

Alex Dos Santos is an employee of Johnson & Johnson and holds stock options.

Introduction

- Apalutamide (APA) combined with continuous androgen deprivation therapy (ADT) produces a rapid and deep prostate-specific antigen (PSA) decline and is associated with improved overall survival and sustained quality of life in participants (pts) with metastatic castration-sensitive prostate cancer (mCSPC)[1,2]
- LIBERTAS (NCT05884398) is a phase 3, prospective, open-label, randomized study evaluating APA with intermittent ADT in pts with mCSPC[3]
- Although ADT is known to affect cognitive domains, such as visuospatial ability and executive function, the association between cognition and treatment responses remains unclear[4]
- We evaluated differences in cognitive performance between PSA responders and nonresponders across visits during the initial treatment phase of LIBERTAS and assessed longitudinal changes in cognition within and between PSA response groups

Methods

Study population

- In LIBERTAS, pts with PSA <0.2 ng/mL at the end of the 6-month initial treatment phase were defined as PSA responders, and all others were defined as PSA nonresponders

Cognitive assessments

- Cognition was assessed via Cambridge Condition's CANTAB battery at baseline (Cycle 1), 2 months (Cycle 3), and 4 months (Cycle 5)
- CANTAB tasks and study procedures were explicitly designed to minimize practice effects. Two cognitive tasks were evaluated (**Table**)

Table. Cognitive Tasks and Measures in the CANTAB Assessment Battery

Cognitive Tasks	Inference	Definition
Paired Associates Learning (PAL) for visual memory	PAL First Attempt Memory Score (PALFAMS28)	Higher values indicate that pts more correctly recalled the location of a visual pattern on their first attempt
	PAL Total Errors Adjusted (PALTEA28)	Lower values indicate fewer number of times that pts chose the incorrect location when recalling a visual pattern[a]
Spatial Working Memory (SWM) for executive function	SWM Between Errors[b] (SWMBE468)	Lower values indicate better working memory[c] performance

[a]The number of incorrect selections was adjusted for estimated errors on uncompleted problems and were comparable across pts regardless of task completion.
[b]"Between Error" is the number of revisits to boxes in which a token had already been found.
[c]Working memory is a cognitive system with limited capacity that temporarily maintains and manipulates information in support of complex tasks such as reasoning, comprehension, and learning.

Statistical analysis

- Cross-sectional differences between PSA responders and nonresponders at each visit were evaluated using the Mann-Whitney U test
- Longitudinal changes within PSA responders and nonresponders were evaluated using linear mixed-effects (LME) models, adjusted for age and baseline testosterone levels
- To assess whether cognitive changes over time differed by treatment response, an additional LME model including a responder status x visit interaction was fitted, adjusting for age and baseline testosterone
- Outlier analysis was not performed to maximize data integrity
- Nominal p values were reported without adjustment for multiplicity

Results

Baseline characteristics (Supplementary Table 1)

- Among 420 pts in the 6-month initial treatment phase, 147 (35.0%) were ADT-naive
- Median (interquartile range) age was 70 (59-81) years in both the overall population and ADT-naive pts

Improved cognitive performance was observed in PSA responders across all visits

- PSA responders demonstrated higher PAL First Attempt Memory Score than nonresponders at each visit (P<0.05; **Figure 1A**)
- PSA responders showed lower PAL Total Errors Adjusted compared with nonresponders at all visits (P<0.05; **Figure 1B**)
- PSA responders exhibited fewer SWM Between Errors than nonresponders at each visit (P<0.05; **Figure 1C**)
- Although practice effects cannot be fully excluded in any serial cognitive testing, the observed patterns in PALFAMS28, PATEA28, and SWMBE468 did not support a classic practice-effect explanation (**Supplementary Figures 1-3**)

Figure 1. Cognitive performance over time by PSA response in all pts

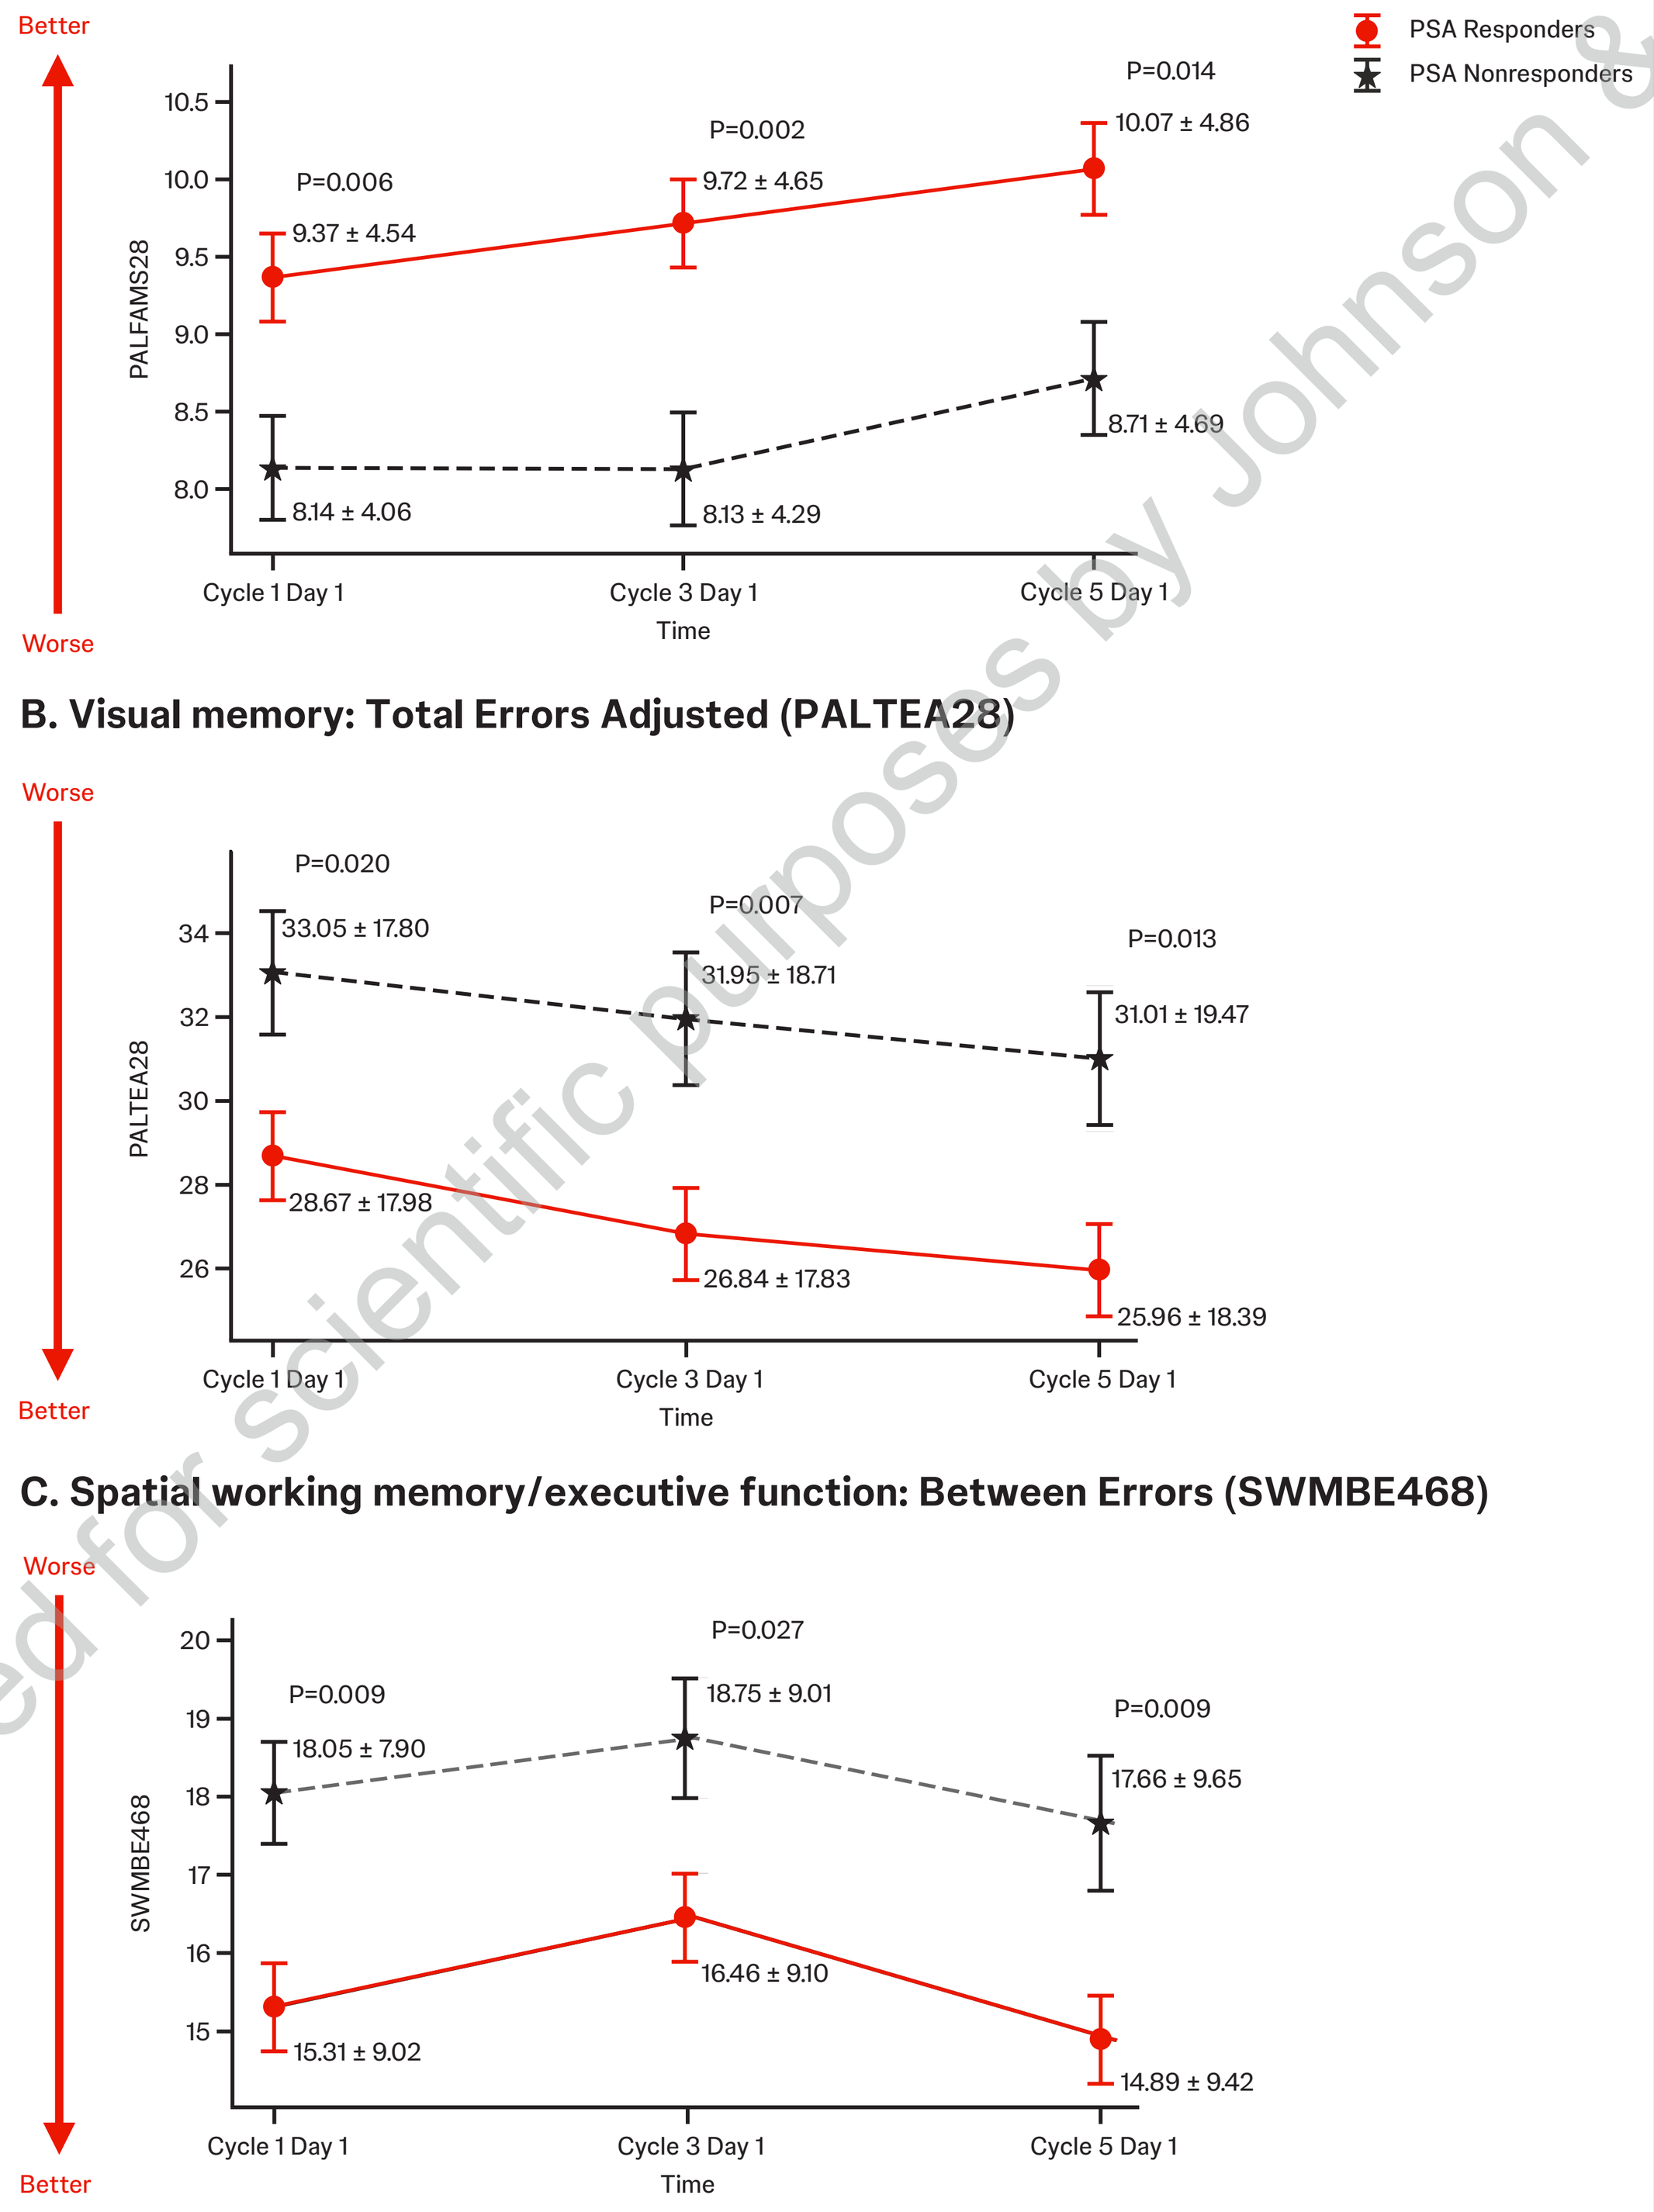
A. Visual memory: First Attempt Memory Score (PALFAMS28)
Better
Worse
PSA Responders
PSA Nonresponders
PALFAMS28
P=0.006
9.37 ± 4.54
8.14 ± 4.06
P=0.002
9.72 ± 4.65
8.13 ± 4.29
P=0.014
10.07 ± 4.86
8.71 ± 4.69
Cycle 1 Day 1
Cycle 3 Day 1
Cycle 5 Day 1
Time
B. Visual memory: Total Errors Adjusted (PALTEA28)
Worse
Better
PALTEA28
P=0.020
33.05 ± 17.80
28.67 ± 17.98
P=0.007
31.95 ± 18.71
26.84 ± 17.83
P=0.013
31.01 ± 19.47
25.96 ± 18.39
Cycle 1 Day 1
Cycle 3 Day 1
Cycle 5 Day 1
Time
C. Spatial working memory/executive function: Between Errors (SWMBE468)
Worse
Better
SWMBE468
P=0.009
18.05 ± 7.90
15.31 ± 9.02
P=0.027
18.75 ± 9.01
16.46 ± 9.10
P=0.009
17.66 ± 9.65
14.89 ± 9.42
Cycle 1 Day 1
Cycle 3 Day 1
Cycle 5 Day 1
Time

Circles and error bars represent mean ± standard error of the mean. Data labels adjacent to each group represent mean ± standard deviation. P-values are calculated using Mann–Whitney U test comparing PSA responders and nonresponders at each visit.

Cognitive advantages in ADT-naive PSA responders

- PSA responders (n=100) had better PAL First Attempt Memory Score than nonresponders (n=44) at 2 months (mean ± SD: 10.45 ± 4.73 vs 8.24 ± 4.28; P=0.01; **Figure 2A**)
- There was no difference in PAL Total Errors Adjusted between PSA responders and nonresponders at any visit in ADT-naive pts (**Figure 2B**)
- Spatial Working Memory for executive function was lower in ADT-naive PSA responders vs ADT-naive PSA nonresponders at baseline (mean ± SD: 14.34 ± 9.12 vs 19.30 ± 8.37; P=0.002) and 4 months (mean ± SD: 15.59 ± 8.62 vs 19.77 ± 11.06; P=0.018; **Figure 2C**)

Figure 2. Cognitive performance over time in ADT-naive pts by PSA response

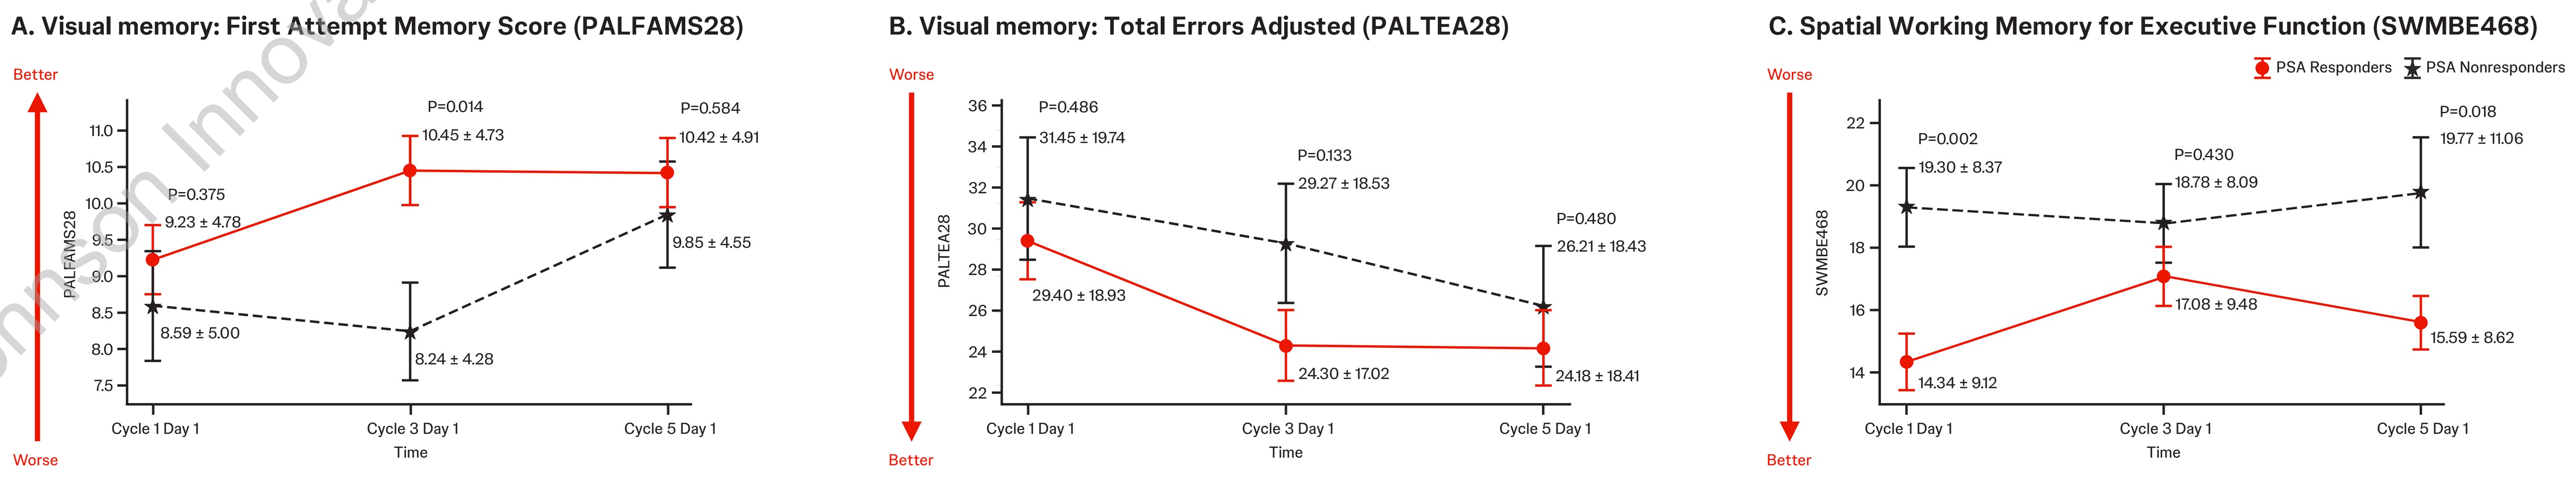
A. Visual memory: First Attempt Memory Score (PALFAMS28)
Better
Worse
PALFAMS28
P=0.375
9.23 ± 4.78
8.59 ± 5.00
P=0.014
10.45 ± 4.73
8.24 ± 4.28
P=0.584
10.42 ± 4.91
9.85 ± 4.55
Cycle 1 Day 1
Cycle 3 Day 1
Cycle 5 Day 1
Time
B. Visual memory: Total Errors Adjusted (PALTEA28)
Worse
Better
PALTEA28
P=0.486
31.45 ± 19.74
29.40 ± 18.93
P=0.133
29.27 ± 18.53
24.30 ± 17.02
P=0.480
26.21 ± 18.43
24.18 ± 18.41
Cycle 1 Day 1
Cycle 3 Day 1
Cycle 5 Day 1
Time
C. Spatial Working Memory for Executive Function (SWMBE468)
PSA Responders
PSA Nonresponders
Worse
Better
SWMBE468
P=0.002
19.30 ± 8.37
14.34 ± 9.12
P=0.430
18.78 ± 8.09
17.08 ± 9.48
P=0.018
19.77 ± 11.06
15.59 ± 8.62
Cycle 1 Day 1
Cycle 3 Day 1
Cycle 5 Day 1
Time

Circles and error bars represent mean ± standard error of the mean. Data labels adjacent to each group represent mean ± standard deviation. P-values are from Mann–Whitney U tests comparing PSA responders and nonresponders at each visit.

PSA responders demonstrated increased accuracy and reduced error of PAL and SWM outcomes over time, whereas nonresponders did not

- Significant cognitive changes over time were observed among PSA responders (P<0.01); no comparable effects were observed with nonresponders (**Figure 3**)
 - Findings were consistent in both the entire sample and the ADT-naive subgroup (**Figure 4**)
- PSA responders showed improved PAL First Attempt Memory Score and reduced PAL Total Errors Adjusted over time
- There was no change in between-group cognitive differences between PSA responders and nonresponders over time in the LME model (P>0.05); cognitive differences between PSA responders and nonresponders remained stable over time (**Supplementary Table 2**)

Figure 3. Longitudinal cognitive changes by PSA response for CANTAB measures in all participants

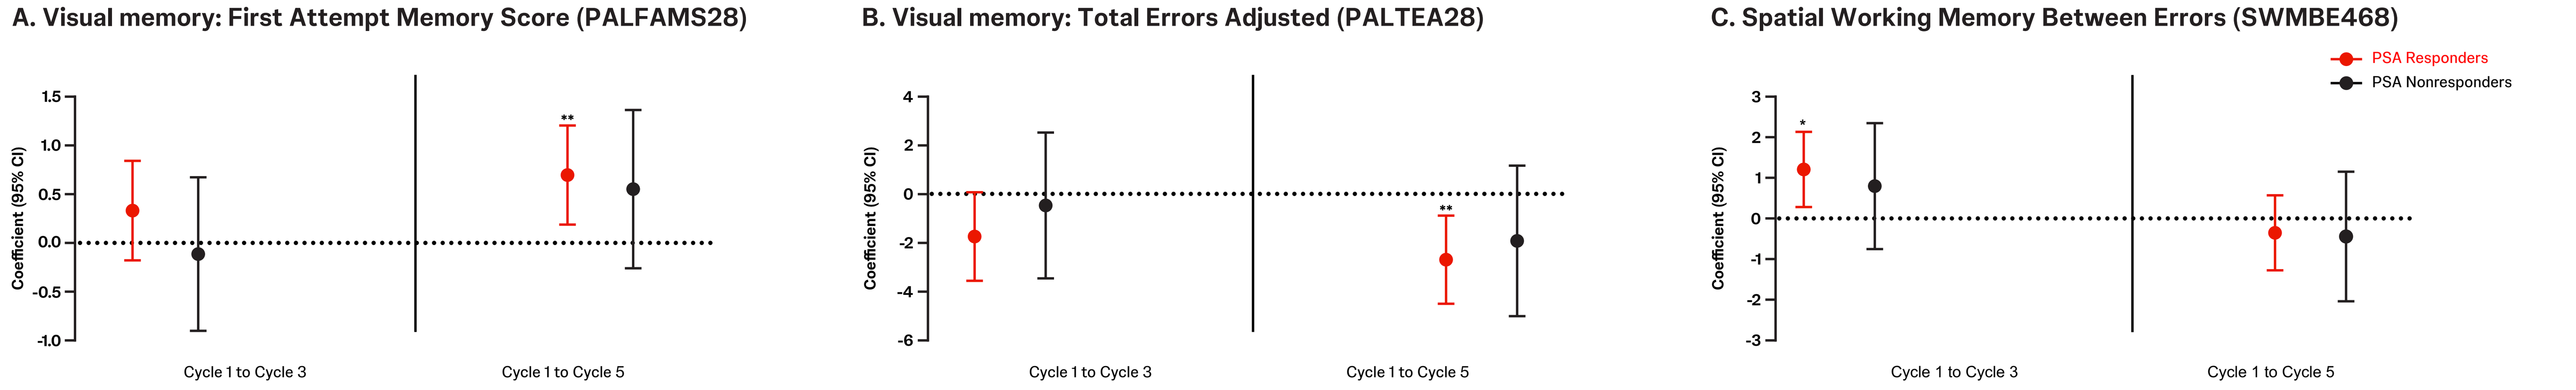
A. Visual memory: First Attempt Memory Score (PALFAMS28)
B. Visual memory: Total Errors Adjusted (PALTEA28)
C. Spatial Working Memory Between Errors (SWMBE468)
PSA Responders
PSA Nonresponders
Coefficient (95% CI)
Cycle 1 to Cycle 3
Cycle 1 to Cycle 5

*P<0.05. **P<0.01.

Figure 4. Longitudinal cognitive changes by PSA response for CANTAB measures in ADT-naive participants

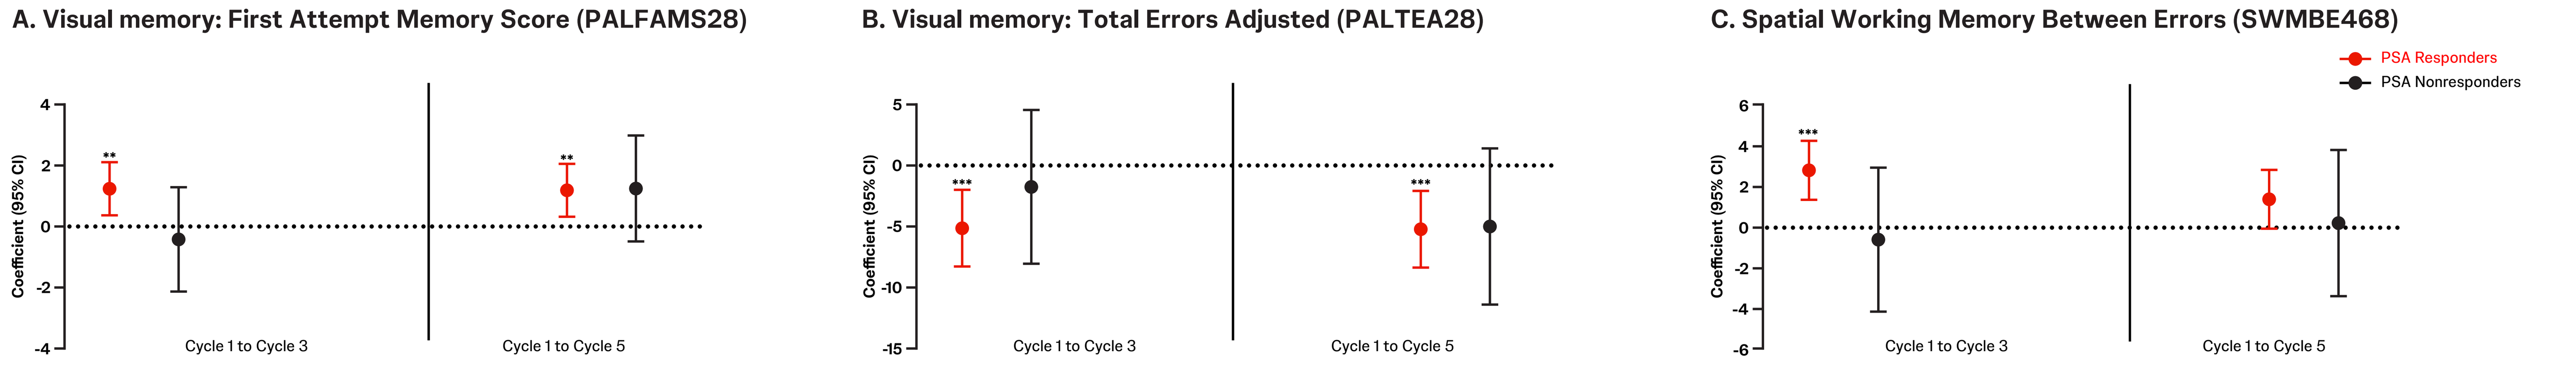
A. Visual memory: First Attempt Memory Score (PALFAMS28)
B. Visual memory: Total Errors Adjusted (PALTEA28)
C. Spatial Working Memory Between Errors (SWMBE468)
PSA Responders
PSA Nonresponders
Coefficient (95% CI)
Cycle 1 to Cycle 3
Cycle 1 to Cycle 5

P<0.01. *P<0.001.

Limitations

- Post-treatment subgroups (eg, PSA responders vs nonresponders at 6 months) may introduce bias but can still offer useful exploratory insights for future hypothesis-driven studies[5,6]

References

1. Chowdhury S, et al. *Ann Oncol* 2023;34(5):477–485. **2.** Small EJ, et al. *Eur Urol Oncol* 2024;7:844–852. **3.** https://clinicaltrials.gov/study/NCT05884398. Accessed April 13, 2026. **4.** Barreira JV, et al. *Cancers (Basel)* 2025;17(15):2501. **5.** Frangakis CE and Rubin DB. *Biometrics* 2002;58 (1): 21–29. **6.** Magnusson BP, et al. *Statistics in Medicine* 2019;38:4761–4771.

Prostate Cancer

Presented by A. Dos Santos at ASCO 2026; May 29–June 2, 2026; Chicago, IL, USA

This material is distributed for scientific purposes by Johnson & Johnson Innovative Medicine and is not for promotional use.