MP37-11

Outcomes, Healthcare Resource Utilization and Costs of Patients Receiving Definitive Versus Non-Definitive Treatment for Muscle-Invasive Bladder Cancer: a Real-World Analysis within the Veterans Affairs System

Renning Zheng[1-2], Jaruda Ithisuphalap[3], Bolan Linghu[4], Eleanor O. Caplan[4], Kirk A Keegan[5-6], Sydney A. McIntire[3], Joshua A. Parrish[3], Claire Evans[3], Amanda M. De Hoedt[3], Sanjay Das[2,7], Ariel B. Bourla[4], Sabine Brookman-May[4,8], Hussein Sweiti[4], Joel Greshock[4], Stephen J. Freedland[2,3,9], Stephen B. Williams[10], Anirban P. Mitra[2,9,11]

School of Medicine, Tsinghua Medicine, Tsinghua University, Beijing, China[1], Department of Urology, Cedars-Sinai Medical Center, Los Angeles, CA[2], Department of Surgery, Durham Veterans Affairs Health Care System, Durham, NC[3], Janssen Research & Development, LLC, Raritan, NJ[4], Nashville Veterans Affairs Health Care System, Nashville, TN[5], Vanderbilt University, Department of Urology, Nashville, TN[6], Cleveland Clinic, Digestive Disease & Surgery Institute, Cleveland OH[7], University of Munich, LMU, Munich, Germany[8], Samuel Oschin Comprehensive Cancer Institute, Cedars-Sinai Medical Center, Los Angeles, CA[9], Department of Surgery, Division of Urology, The University of Texas Medical Branch, Galveston, TX[10], Department of Computational Biomedicine, Cedars-Sinai Medical Center, Los Angeles, CA[11]

INTRODUCTION

- Approximately 50% of patients with muscle-invasive bladder cancer (MIBC) do not receive definitive treatments (DT), including radical cystectomy (RC) or chemoradiation (CRT), despite established survival benefits[1-4].
- Retrospective real-world data were used to better understand and compare demographic, clinical characteristics, survival outcomes, and healthcare resource utilization (HCRU) and associated costs among patients with MIBC receiving DT vs. no definitive treatment (NDT).

METHODS

- Patients from the United States Veterans Affairs (VA) System diagnosed with MIBC between 2010-2019 were identified via a validated natural language processing (NLP) model analyzing pathology reports of transurethral resection of bladder tumor (TURBT) and supplementary chart review[5].
- Index date was defined as the first date of MIBC diagnosis. All eligible patients had baseline periods of ≥12 months pre-index and follow-up periods of >3 months post-index.
- Baseline characteristics were summarized and compared by omnibus (i.e., global) tests.
- The impact of treatment groups on overall survival (OS) was visualized by Kaplan-Meier plot and tested by a multivariable Cox model with time-dependent covariates. Competing risk model was used for BC-specific survival to account for death from other causes and visualized with cumulative incidence function curves.
- All-cause and bladder-cancer specific (BC-specific) HCRU and associated costs were summarized.

Urothelial Cancer

RESULTS

- We identified **1,524 patients with MIBC**, of whom **650 received NDT**, **740 were treated with RC**, and **134 were treated with CRT**.
- **Patients who received NDT were older at MIBC diagnosis** (median age in years: 78 [NDT], 68 [RC], 72 [CRT]; p<0.001).

- An **OS advantage** was observed for **patients receiving DT vs. NDT** in both **unadjusted** Kaplan-Meier plot and the **adjusted** multivariable Cox model (adjusted hazard ratios (HR): 0.49 [RC], 0.65 [CRT]; both, p<0.001) **(Figures 1 and 2)**

FIGURE 1: Unadjusted Kaplan-Meier plot of OS

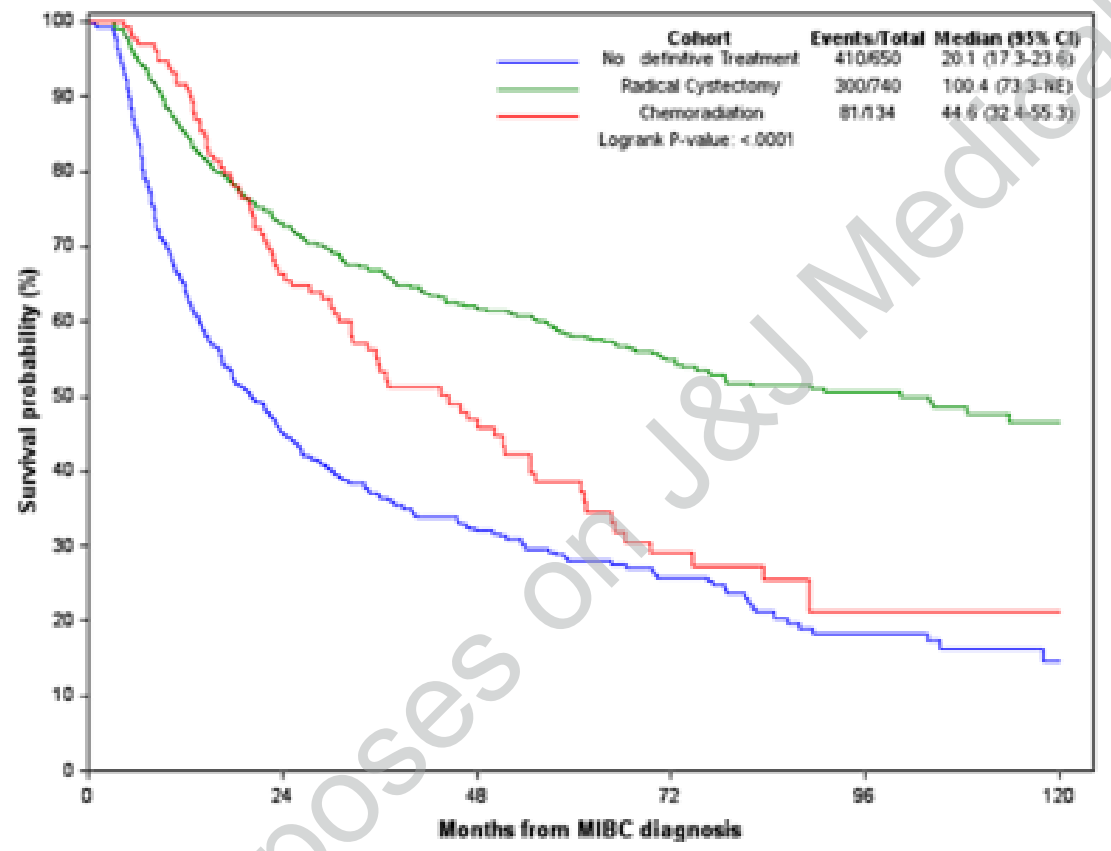
Cohort
Events/Total
Median (95% CI)
No definitive Treatment 410/650 20.1 (17.3-23.6)
Radical Cystectomy 300/740 100.4 (73.3-NE)
Chemoradiation 81/134 44.6 (32.4-55.3)
Logrank P-value: <.0001
Survival probability (%)
Months from MIBC diagnosis

FIGURE 2: Adjusted Kaplan-Meier plot of OS

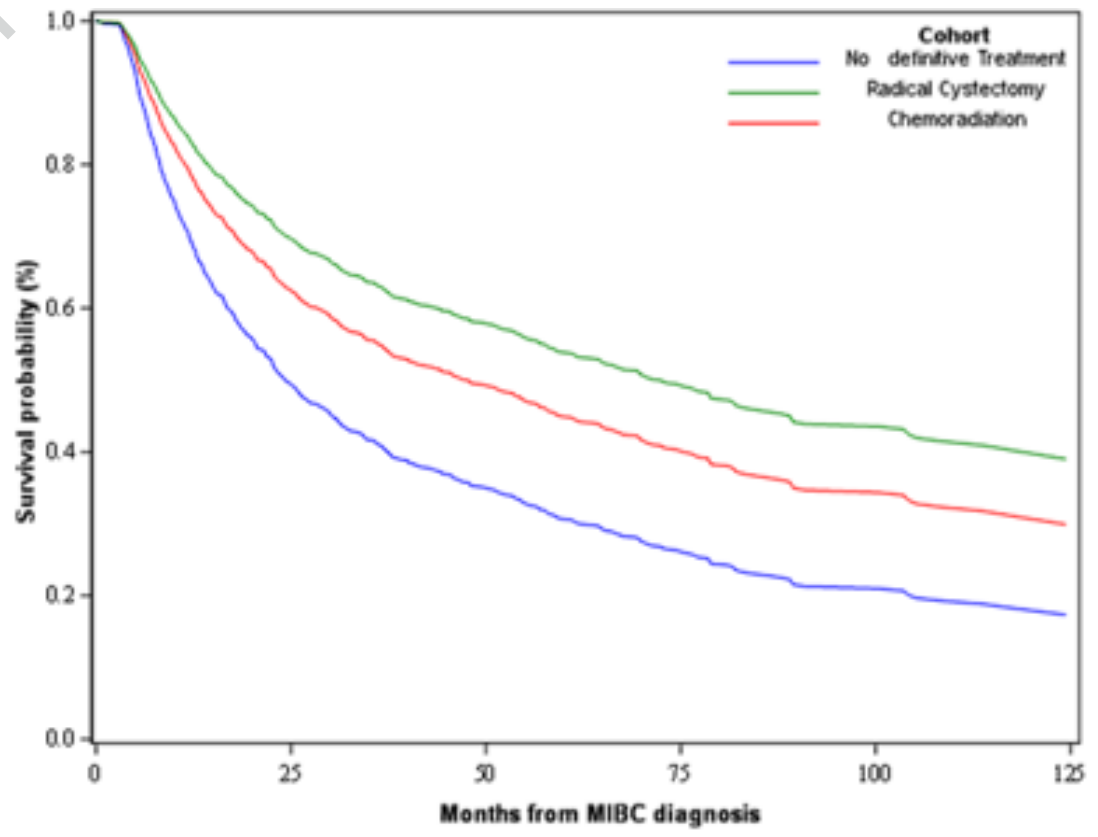
Cohort
No definitive Treatment
Radical Cystectomy
Chemoradiation
Survival probability (%)
Months from MIBC diagnosis

- The probability **of BC-specific mortality**, accounting for the probability of mortality due to other causes, was **higher** over time **among those with NDT vs. DT** in both **unadjusted** and **adjusted** models **(Figures 3 and 4)**.
Results in 2 and 4 were adjusted for age, year of diagnosis, race, smoking status, Charlson Comorbidity Index (CCI), proportion living below poverty line in zip code, region, presence of carcinoma in situ (CIS) and history of intravesical Bacillus Calmette-Guerin (BCG) and/or chemotherapy.

FIGURE 3: Unadjusted Cumulative incidence plot of BC-specific mortality

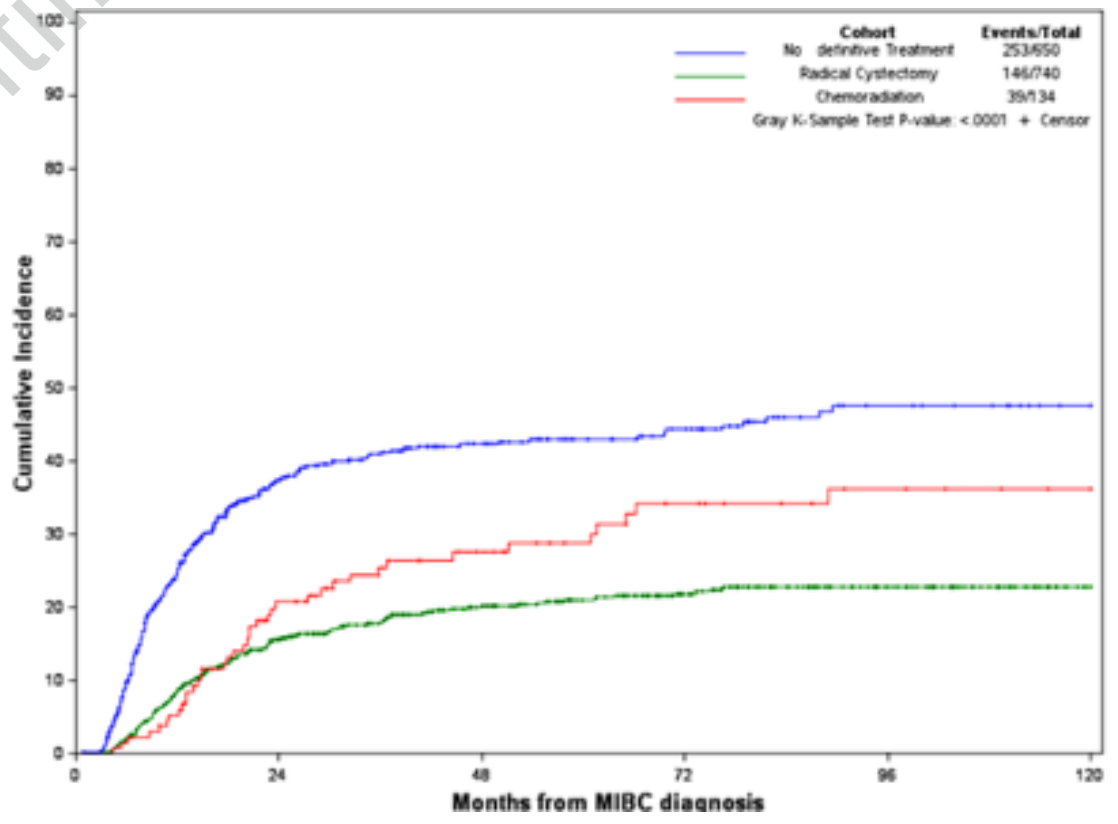
Cohort
Events/Total
No definitive Treatment 253/650
Radical Cystectomy 146/740
Chemoradiation 39/134
Gray K-Sample Test P-value: <.0001 + Censor
Cumulative Incidence
Months from MIBC diagnosis

FIGURE 4: Adjusted Cumulative incidence plot of BC-specific mortality

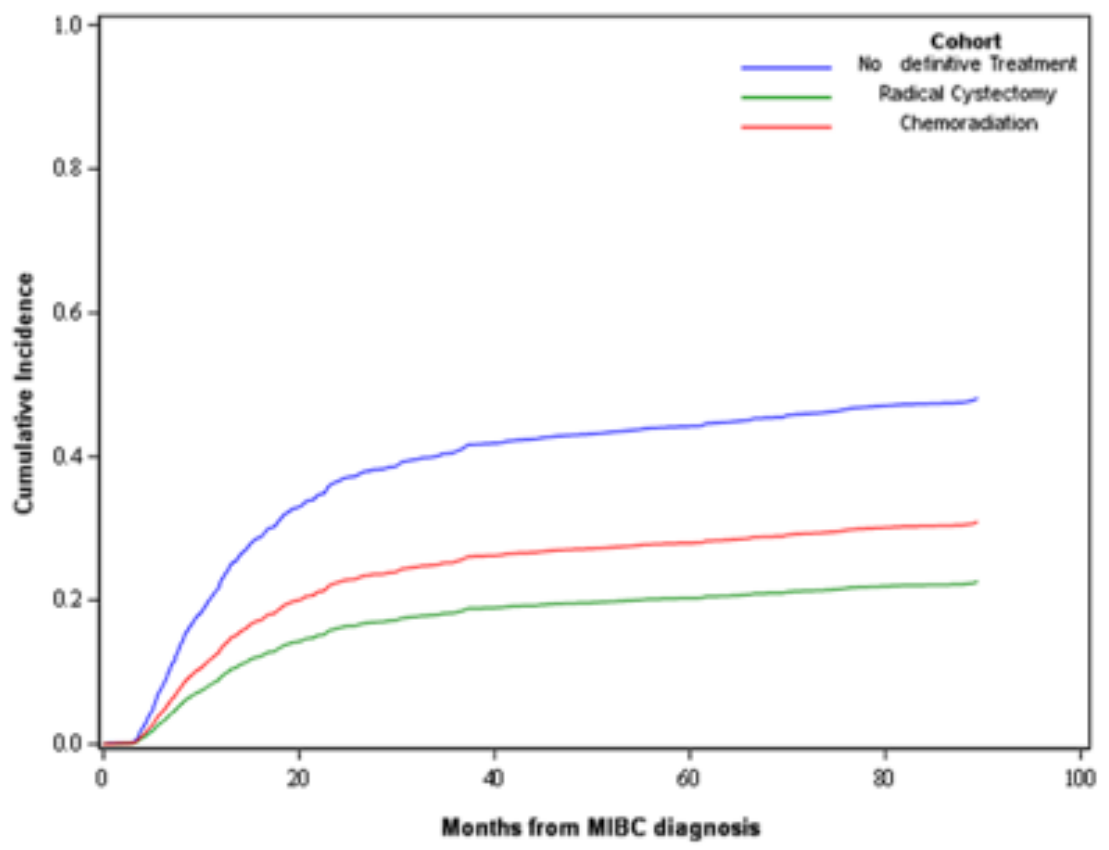
Cohort
No definitive Treatment
Radical Cystectomy
Chemoradiation
Cumulative Incidence
Months from MIBC diagnosis

- Inpatient HCRU and costs were highest for patients receiving RC. Outpatient HCRU and costs were highest for patients receiving CRT.

REFERENCES:

1. Patel M, Gadzinski AJ, Bell AM, et al. Interprofessional Consultations (eConsults) in Urology. *Urol Pract.* May 2021;8(3):321-327.
2. Westergren DO, Gårdmark T, Lindhagen L, Chau A, Malmström PU. A Nationwide, Population Based Analysis of Patients with Organ Confined, Muscle Invasive Bladder Cancer Not Receiving Curative Intent Therapy in Sweden from 1997 to 2014. *J Urol.* Nov 2019;202(5):905-912.
3. Gray PJ, Fedewa SA, Shipley WU, et al. Use of potentially curative therapies for muscle-invasive bladder cancer in the United States: results from the National Cancer Data Base. *Eur Urol.* May 2013;63(5):823-829.
4. ohn JB, Varughese MA, Cooper N, et al. Treatment Allocation and Survival in Patients Diagnosed with Nonmetastatic Muscle-invasive Bladder Cancer: An Analysis of a National Patient Cohort in England. *Eur Urol Focus.* Mar 2021;7(2):359-365.
5. Yang R, Zhu D, Howard LE, et al. Context-Based Identification of Muscle Invasion Status in Patients With Bladder Cancer Using Natural Language Processing. *JCO Clin Cancer Inform.* Jan 2022;6:e2100097.

Presented by Renning Zheng at AUA 2025 Annual Meeting; April 2025; Las Vegas, NV, USA

KEY TAKEAWAY

Patients receiving NDT were older, had significantly worse survival, and incurred lower but still substantial HCRU compared to those receiving DT.

CONCLUSIONS

- Patients receiving NDT were older and had worse overall survival compared to those receiving DT.
- These results within the VA may not reflect those in other systems or populations (e.g., Medicare, Commercial) or costs within specific timeframes.
- Further studies are needed to evaluate other impacts of DT and NDT on patients including productive life-years lost and the physical and emotional burden.
- These findings highlight unmet needs among MIBC patients receiving NDT and underscore the requirement for more tolerable novel therapies to improve the prognosis of these patients.

ACKNOWLEDGMENTS

This study was conducted with the financial support of Janssen Research & Development, LLC.

DISCLOSURES

RZ and SD have no disclosures to report. BL, RH, EOC, JG report employment with J&J. KK is a shareholder in J&J and CG Oncology. HS employment with J&J and ownership of J&J stocks. SB owns J&J stocks and was a J&J employee at the time of study. SJF consultant to Janssen, Astellas, Bayer, Pfizer, Sanofi, Sumitomo, Novartis, Astra Zeneca, Merck, and Eli Lilly. SBW reports advisory board and consultant fees from Janssen, Merck, Photocure, Valar Labs and serves as BJUI section editor. APM reports honoraria from UpToDate and is a co-creator of intellectual property owned by the University of Southern California related to a prognostic panel for urinary bladder cancer. JI, SM, JP, CE, AD report research funding paid to the lab by: Merck, Janssen, Exact Sciences, Astellas, Delfi Diagnostics, Guardant Health, Photocure, AstraZeneca Pharmaceuticals, Reinvestment Partners, Prostatype Genomics, Novartis, Vir Biotechnology, Coloplast corporation, Roche Pharmaceuticals

Scan the QR code

https://www.congresshub.com/Oncology/AUA2025/TAR-200/Zheng

The QR code is intended to provide scientific information for individual reference, and the information should not be altered or reproduced in any way.

This material is distributed for scientific purposes on J&J Medical Connect, and is not for promotional use