304b

OrigAMI-3: A randomized, phase 3 study of amivantamab plus FOLFIRI vs cetuximab or bevacizumab plus FOLFIRI in participants with recurrent, unresectable, or metastatic *RAS/BRAF* wild-type colorectal cancer

J Randolph Hecht[1,*], Elena Élez[2], Cathy Eng[3], Peter Gibbs[4], Jenny Seligmann[5], Ruihua Xu[6], Kensei Yamaguchi[7], Jaw Yuan Wang[8], Hao Wei Teng[9], Leonardo Trani[10], Honeylet Wortman-Vayn[11], Zhengyu Jiang[10], Brooke Diorio[12], Patricia Lorenzini[12], Christine Baudelet[13], Seema Sethi[10], Mahadi Baig[12], Chiara Cremolini[14]

[1]UCLA Jonsson Comprehensive Cancer Center, Santa Monica, CA, USA; [2]Vall d'Hebron University Hospital, Barcelona, Spain; [3]Vanderbilt-Ingram Cancer Center, Nashville, TN, USA; [4]Western Health Sunshine Hospital, St Albans, VIC, Australia; [5]St James University Hospital, Leeds, UK; [6]Sun Yat-Sen University Cancer Center, Guangzhou, China; [7]The Cancer Institute Hospital of JFCR, Tokyo, Japan; [8]Kaohsiung Medical University Chung Ho Memorial Hospital, Kaohsiung Medical University, Kaohsiung, Taiwan; [9]Taipei Veterans General Hospital, Taipei, Taiwan; [10]Johnson & Johnson, Spring House, PA, USA; [11]Johnson & Johnson, Bridgewater, NJ, USA; [12]Johnson & Johnson, Raritan, NJ, USA; [13]Janssen Research & Development, a Johnson & Johnson company, Beerse, Belgium; [14]Azienda Ospedaliero Universitaria Pisana, Pisa, Italy

*Presenting author: jrhecht@mednet.ucla.edu.

Background

- Approximately 50% of patients with metastatic colorectal cancer (CRC) have tumors that are wild-type (WT) for *KRAS*, *NRAS*, and *BRAF* (*RAS/BRAF* WT) without actionable genomic alterations[1]
- Standard first-line (1L) therapy for *RAS/BRAF* WT metastatic CRC is 5-fluorouracil–based doublet chemotherapy (FOLFOX or FOLFIRI) plus anti–epidermal growth factor receptor (EGFR) or anti–vascular endothelial growth factor (VEGF) therapy[2] with second-line (2L) treatment being dependent on 1L treatment; however, resistance is nearly inevitable[3,4]
 - Clinical outcomes for metastatic CRC are poor, with 5-year survival rates in the United States of <20%[5]
 - In 2L RAS WT metastatic CRC, cetuximab plus irinotecan demonstrated a median progression-free survival of 5.4 months and a median overall survival of 12.3 months[6]
- *MET* alterations are known resistance mechanisms to anti-EGFR therapies and increase in prevalence with subsequent lines of therapy[4,7,8]
- Amivantamab, an EGFR-MET bispecific antibody with immune cell–directing activity (**Figure 1**),[9] is US Food and Drug Administration approved for 4 indications in *EGFR*-mutated advanced non-small cell lung cancer[10]
- In the phase 1b/2 OrigAMI-1 study (ClinicalTrials.gov Identifier: NCT05379595), amivantamab plus FOLFOX or FOLFIRI demonstrated promising antitumor activity, regardless of line of therapy or tumor sidedness, in participants with *RAS/BRAF* WT metastatic CRC without prior anti-EGFR exposure[11]
 - Objective response rate was 44% (14/32 participants [complete response, 1; partial response, 13; unconfirmed partial response, 1; stable disease, 13; progressive disease, 4]) among participants with *RAS/BRAF* WT metastatic CRC who received 2L amivantamab plus FOLFOX or FOLFIRI, with several being able to proceed with curative-intent surgery[11]

FIGURE 1: Amivantamab's triple mechanism of action

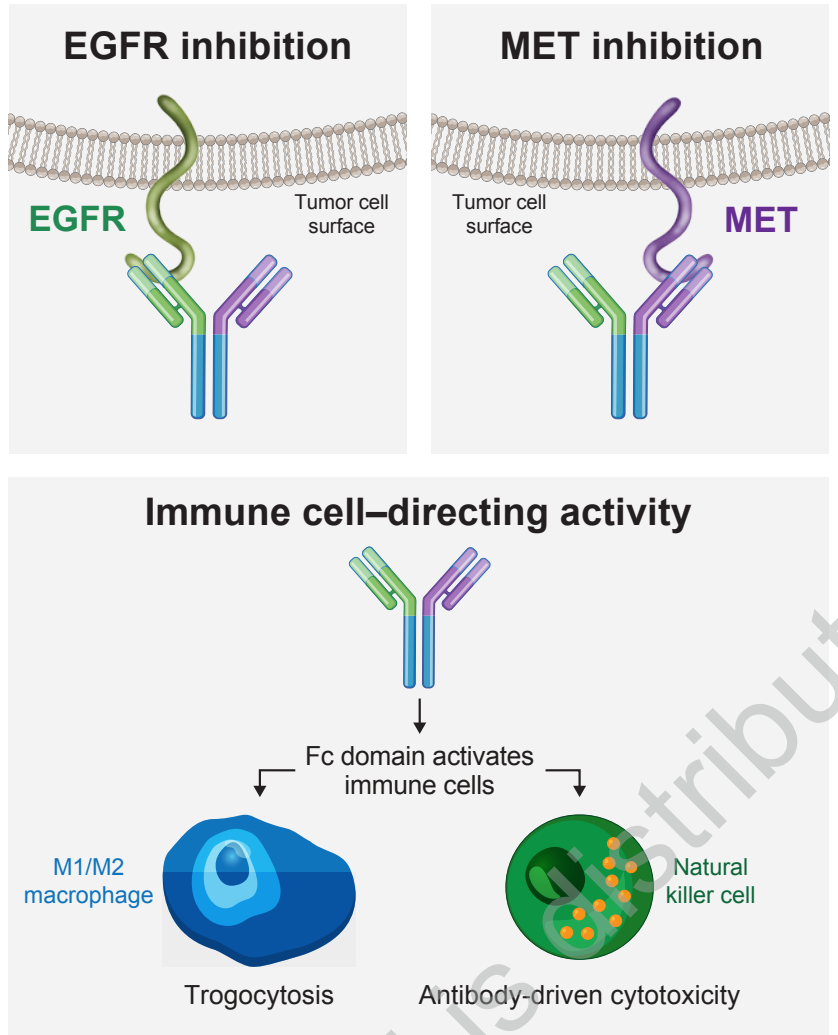
EGFR inhibition
MET inhibition
EGFR
Tumor cell surface
Tumor cell surface
MET
Immune cell–directing activity
Fc domain activates immune cells
M1/M2 macrophage
Natural killer cell
Trogocytosis
Antibody-driven cytotoxicity

EGFR, epidermal growth factor receptor.

Objective

- The phase 3, randomized OrigAMI-3 study will evaluate the efficacy and safety of subcutaneous (SC) amivantamab (co-formulated with recombinant human hyaluronidase PH20) plus FOLFIRI compared with intravenous (IV) cetuximab or bevacizumab plus FOLFIRI as 2L therapy for participants with *RAS/BRAF* WT recurrent, unresectable, or metastatic CRC

Methods

- OrigAMI-3 is a randomized, open-label, phase 3 study (ClinicalTrials.gov Identifier: NCT06750094) currently recruiting participants with recurrent, unresectable, or metastatic CRC (**Figure 2**)
- Participants must have disease progression on 1 prior line of systemic therapy for metastatic disease
 - Prior regimen must be fluoropyrimidine- and oxaliplatin-based therapy, with or without anti-VEGF treatment
- Participants will be randomized 1:1 to receive amivantamab SC or anti-EGFR/anti-VEGF therapy (cetuximab IV or bevacizumab IV; per investigator's choice prior to randomization), both in combination with chemotherapy (FOLFIRI)
 - Recruitment for cetuximab IV or bevacizumab IV will be capped to ensure a sufficient number of participants receive either treatment
- Participants may undergo curative-intent surgery/procedure if appropriate during the treatment period
 - All cases will be reviewed by an independent committee prior to the intervention

FIGURE 2: OrigAMI-3 study design

Key eligibility criteria
- Recurrent, unresectable, or metastatic CRC
- Received 1 line of systemic therapy for metastatic CRC
- Anti-EGFR therapy and irinotecan naïve
- *KRAS*, *NRAS*, and *BRAF* WT tumor as determined by local testing
- ECOG PS score of 0 or 1

Stratification factors
- Investigator's choice of control prior to randomization (cetuximab/bevacizumab)
- Primary tumor location (left/right)[a]
- Prior anti-VEGF therapy (yes/no)
- Duration of 1L therapy[b] (<6 mo/≥6 mo)

1:1 randomization (N≈700)

R

Amivantamab SC[c] + FOLFIRI (n≈350)

Cetuximab IV or Bevacizumab IV + FOLFIRI (n≈350)

Dosing (in 28-day cycles)
Amivantamab: 1600 mg SC (2240 mg if body weight ≥80 kg)
Cycle 1: Weekly administration (Days 1, 8, 15, and 22)
Cycle 2+: Biweekly administration (Days 1 and 15)
Cetuximab: Biweekly IV regimen (500 mg/m² on Days 1 and 15) or weekly IV regimen (400 mg/m² on C1D1, then 250 mg/m² weekly)
Bevacizumab: 5 mg/kg IV on Days 1 and 15

Dual primary endpoints:
- PFS per RECIST v1.1 by BICR
- OS

Secondary endpoints:
- PFS by investigator
- ORR, DoR, DCR (by BICR and investigator)
- PFS after first subsequent therapy (by investigator)
- TTF
- Curative resection rate[d]
- AEs assessed via NCI-CTCAE v5.0 and laboratory abnormalities
- PROs assessed via EORTC QLQ-C30, EORTC QLQ-CR29, EQ-5D-5L, and EORTC Q168

[a]Left-sided CRC is defined as a primary tumor that arises from the splenic flexure, descending colon, sigmoid colon, rectosigmoid, or rectum; right-sided CRC is defined as a primary tumor that arises from the cecum, ascending colon, or transverse colon. [b]Start of therapy to disease progression. [c]Co-formulated with 2000 U/mL rHuPH20. [d]For participants who undergo curative-intent surgery/procedure during the treatment period, preintervention imaging will be used to determine the best response of CR, PR, or SD, and postintervention tumor assessments will be used to determine the time and location of PD (eg, recurrence at the site of resection or a distant new lesion).
1L, first-line; AE, adverse event; BICR, blinded independent central review; C, Cycle; CR, complete response; CRC, colorectal cancer; D, Day; DCR, disease control rate; DoR, duration of response; ECOG PS, Eastern Cooperative Oncology Group performance status; EGFR, epidermal growth factor receptor; EORTC QLQ, European Organisation for Research and Treatment of Cancer Quality of Life Questionnaire; IV, intravenous; NCI-CTCAE v5.0, National Cancer Institute Common Terminology Criteria for Adverse Events v5.0; ORR, objective response rate; OS, overall survival; PD, progressive disease; PFS, progression-free survival; PR, partial response; PRO, patient-reported outcome; RECIST v1.1, Response Evaluation Criteria in Solid Tumors v1.1; rHuPH20, recombinant human hyaluronidase PH20; SC, subcutaneous; SD, stable disease; TTF, time to treatment failure; VEGF, vascular endothelial growth factor; WT, wild-type.

- Key inclusion and exclusion criteria are shown in **Table 1**

TABLE 1: Key inclusion and exclusion criteria

Inclusion criteria	Exclusion criteria
• ≥18 years of age	• History of or current ILD, pneumonitis, or pulmonary fibrosis
• Recurrent, unresectable, or metastatic CRC	• Known allergies, hypersensitivity, or intolerance to any component of the study treatment
• *KRAS*, *NRAS*, and *BRAF* WT tumor as determined by local testing	• Known dMMR/MSI-H status without prior immunotherapy
• Consent to the submission of fresh or archival tumor tissue	• HER2-positive/amplified tumor
• Received 1 line of systemic therapy for metastatic CRC	• Prior exposure to irinotecan and/or agents that target EGFR or MET
• ECOG PS score of 0 or 1	• History or known presence of leptomeningeal disease or spinal cord compression
• May have brain metastases if definitively and locally treated, clinically stable, and asymptomatic for ≥2 weeks	• Active hepatitis of infectious origin

CRC, colorectal cancer; dMMR, mismatch repair deficiency; ECOG PS, Eastern Cooperative Oncology Group performance status; EGFR, epidermal growth factor receptor; ILD, interstitial lung disease; MSI-H, microsatellite instability–high; WT, wild-type.

OrigAMI-3 enrollment sites

- The multicenter, global OrigAMI-3 study is planned to open at ~230 sites in 26 locations (**Figure 3**)

FIGURE 3: OrigAMI-3 enrollment sites

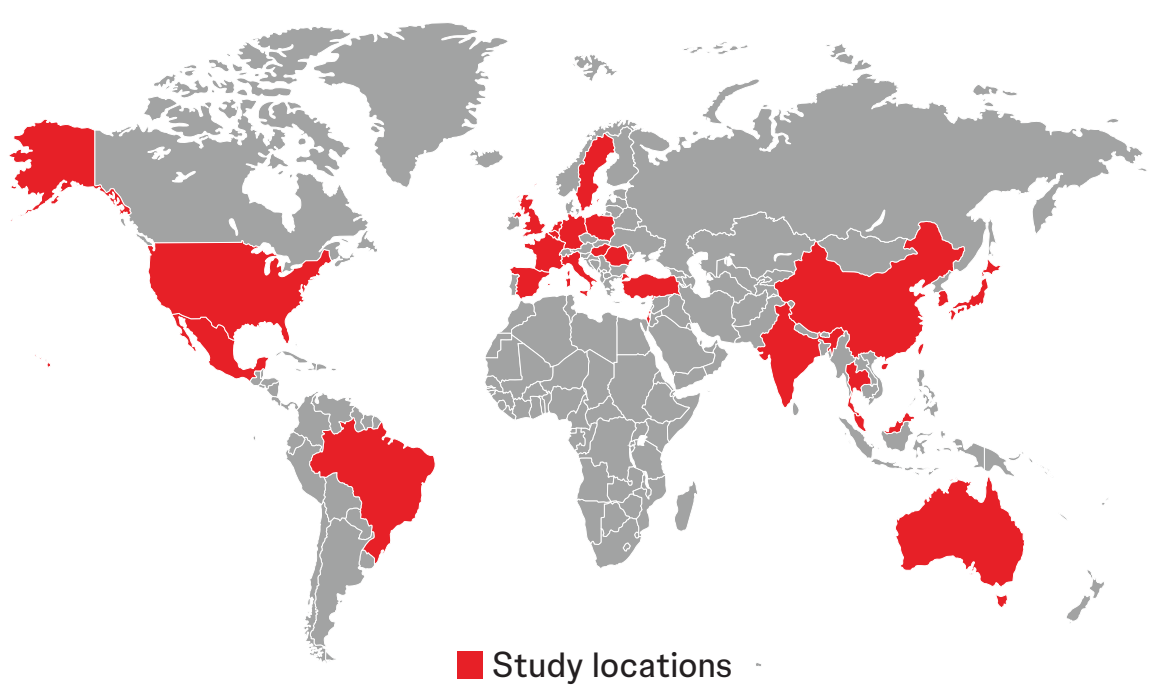
Study locations

Summary

OrigAMI-3 is a randomized, open-label, phase 3 study evaluating the efficacy of amivantamab SC plus FOLFIRI compared with cetuximab IV or bevacizumab IV plus FOLFIRI as 2L treatment for participants with *RAS/BRAF* WT recurrent, unresectable, or metastatic CRC who have received prior chemotherapy

Current status

OrigAMI-3 is currently enrolling, with a goal of 700 participants

Registration information

This study is registered with ClinicalTrials.gov (Identifier: NCT06750094)

Acknowledgments

We thank the individuals who participated in this study and their families and caregivers, the physicians and nurses who cared for the participants, the staff members who supported this clinical trial, and the staff members at the study sites and those who were involved in data collection/analyses. Medical writing and editorial support were provided by Lumanity Communications Inc. and funded by Johnson & Johnson.

Disclosures

JRH received consulting fees from Astellas, Bristol Myers Squibb, Taiho, BeiGene, IGM Biosciences, Novartis, Galvanize, Exelixis, and Deciphera; received payment or honoraria from Scripps, the American Gastroenterological Association, MJH Life Sciences, and From Research to Practice; and owns stock in Triumvira, Actym, and MBQ Pharma. **EÉ** served in a consulting or advisory role for Amgen, Bayer, Boehringer Ingelheim, Bristol Myers Squibb, Cureteq AG, Janssen, Merck, Merck Serono, Merck Sharp & Dohme, Novartis, Pfizer, Pierre Fabre, Repare Therapeutics, RIN Institute Inc., Roche, Sanofi, Seagen, Servier, and Takeda; received honoraria from Amgen, Bayer, Boehringer Ingelheim, Bristol Myers Squibb, Cureteq AG, Janssen, Lilly, Medscape, Merck, Merck Serono, Merck Sharp & Dohme, Novartis, Pfizer, Pierre Fabre, Repare Therapeutics, RIN Institute Inc., Roche, Sanofi, Seagen, Servier, and Takeda; received travel, accommodations, and expenses covered by Amgen, Bayer, Boehringer Ingelheim, Bristol Myers Squibb, Cureteq AG, Janssen, Lilly, Medscape, Merck, Merck Serono, Merck Sharp & Dohme, Novartis, Pfizer, Pierre Fabre, Repare Therapeutics, RIN Institute Inc., Roche, Sanofi, Seagen, Servier, and Takeda; and received research funding from AbbVie, Amgen, Array BioPharma, AstraZeneca, Bayer, BeiGene, Bioncotech, BioNTech RNA Pharmaceuticals GMBH, BioNTech Small Molecules GMBH, Boehringer Ingelheim, Boehringer Ingelheim Spain, Bristol Myers Squibb, Celgene, Daiichi Sankyo, Debiopharm Group, Genentech, GERCOR, HalioDx, F Hoffmann–La Roche, Hutchison MediPharma, Iovance Biotherapeutics, Janssen Research & Development, Janssen-Cilag SA, MedImmune, Menarini, Merck, Merck Sharp & Dohme de España SA, Merus NV, Mirati Therapeutics, Nouscom SRL, Novartis, Novartis Farmacéutica SA, Pfizer, PharmaMar, Pierre Fabre, PledPharma, Redx Pharma, Roche, Sanofi, Scandion Oncology, Seagen, Servier, SOTIO, Taiho Pharma USA, Taiho Pharmaceutical, and WNTresearch. **CE** served in a consulting or advisory role for Elevar Therapeutics, Elevation Oncology, General Electric, GSK, Gritstone Bio, Hutchison MediPharma, Janssen Oncology, Merck, Merck Serono, Natera, Pfizer, and Seagen. **PG** received honoraria from Amgen, Merck, MSD Oncology, Roche, and Servier; served in a consulting or advisory role for Haystack Oncology; received research funding from Amgen, Bayer, Bristol Myers Squibb/Sanofi, Merck Serono, Pierre Fabre, Roche, Roche Molecular Diagnostics, Servier, and Takeda; and received travel, accommodations, and expenses covered by Servier. **JS** served in a consulting or advisory role for Elevation Oncology, GSK, Jazz Pharmaceuticals, Merck Serono, Sanofi, and Takeda; received honoraria from GSK, Merck Serono, Pierre Fabre, Servier, and Takeda Science Foundation; and received research funding from GSK, Merck Serono, and Pierre Fabre. **RX** served in a consulting or advisory role for Astellas, AstraZeneca, BeiGene, CPPC, Hengrui Pharma, Hutchison MediPharma, Innovent Biologics, Junshi Biosciences, Keymed Bioscience, Merck Serono, Merck Sharp & Dohme, Qilu Pharmaceutical, and Roche. **KY** served in a consulting or advisory role for Bristol Myers Squibb Japan and Daiichi Sankyo; participated in a speakers bureau for Chugai Pharma, Bristol Myers Squibb Japan, Takeda, Taiho Pharmaceutical, Lilly, Ono Pharmaceutical, Daiichi Sankyo, and Merck; and received research funding from Ono Pharmaceutical, Taiho Pharmaceutical, Daiichi Sankyo, Lilly, Gilead Sciences, Yakult Honsha, Chugai Pharma, Boehringer Ingelheim, Eisai, MSD Oncology, Sanofi, and Bristol Myers Squibb. **HWT** served in a consulting or advisory role for Merck Sharp & Dohme; participated in a speakers bureau for Merck Sharp & Dohme; and received research funding from Merck Sharp & Dohme. **LT**, **HW-V**, **ZJ**, **BD**, **PL**, **CB**, **SS**, and **MB** are employees of and may hold stock in Johnson & Johnson. **CC** received honoraria from Roche, Amgen, Bayer, Servier, Merck Sharp & Dohme, Merck, and Pierre Fabre; served in a consulting or advisory role for Roche, Bayer, Amgen, Merck Sharp & Dohme, Pierre Fabre, and Nordic Bioscience; participated in a speakers bureau for Servier, Merck, and Pierre Fabre; and received research funding from Merck, Bayer, Roche, and Servier. **JYW** has nothing to disclose.

Metastatic Colorectal Cancer

Scan the QR code

https://www.congresshub.com/Oncology/AM2025/Amivantamab/Hecht

Copies of this poster obtained through Quick Response (QR) Code are for personal use only and may not be reproduced without permission from ASCO® or the authors of this poster.

Presented by JR Hecht at the American Society of Clinical Oncology (ASCO) Annual Meeting; May 30–June 3, 2025; Chicago, IL, USA.

REFERENCES:

1. Biller LH, Schrag D. *JAMA*. 2021;325(7):669–685. 2. Cervantes A, et al. *Ann Oncol*. 2023;34(1):10–32. 3. Misale S, et al. *Nature*. 2012;486(7404):532–536. 4. Sforza V, et al. *World J Gastroenterol*. 2016;22(28):6345–6361. 5. National Cancer Institute SEER Program. Cancer stat facts: colorectal cancer. Accessed April 25, 2025. https://seer.cancer.gov/statfacts/html/colorect.html. 6. Sobrero A, et al. *Oncologist*. 2021;26(2):e261–e269. 7. Raghav K, et al. *Oncotarget*. 2016;7(34):54627–54631. 8. Bray SM, et al. *Sci Rep*. 2019;9(1):15365. 9. Moores SL, et al. *Cancer Res*. 2016;76(13):3942–3953. 10. RYBREVANT® (amivantamab-vmjw) injection for intravenous use [package insert]. Janssen Biotech, Inc.; 2025. 11. Pietrantonio F, et al. Presented at: European Society for Medical Oncology (ESMO) Annual Meeting; September 13–17, 2024; Barcelona, Spain.

This material is distributed for scientific purposes on J&J Medical Connect, and is not for promotional use