Poster Number: 105

Understanding the Assessment and Management of Insomnia Symptoms in Patients With Major Depressive Disorder Using Patient-Provider Conversations From the Verilogue Database

Hrishikesh Kale,[1] Zachary N. Hebert,[2] Samantha Creel,[2] Hank Zhang,[1] Karen Johnston[1]

[1]Janssen Scientific Affairs, LLC, a Johnson & Johnson Company, Titusville, NJ, USA; [2]Verilogue, Philadelphia, PA, USA

This material is distributed for scientific purposes by Johnson & Johnson Innovative Medicine and is not for promotional use.

Background

- The estimated prevalence of a major depressive episode among adults aged 18 or older in the United States for 2021 was 21 million adults or ~8% of all US adults[1]
- As a common symptom of MDD, insomnia[2-4] is associated with:
 - Poorer remission outcomes and higher relapse rates[5]
 - Increased healthcare resource use[6]
 - Impaired health-related quality of life[2]
- While insomnia is one of the key symptoms of MDD[7,8], little is known about how often HCPs and patients mention or discuss insomnia during clinical visits

Objective

- Characterize discussions of insomnia, related symptoms, and treatments among adults with MDD and their psychiatrists/PCPs

Methods

- Dialogues gathered from Verilogue's HCP/patient conversation database were used for a retrospective analysis
 - The database consisted of >238,000 US, anonymized, and syndicated recordings since 2006, including >1700 HCPs and >211,000 patients; recordings span community, academic, hospital, and telehealth settings
 - For this study, eligible visits were recorded in community-based settings (US only) between January 1, 2022, and February 28, 2026 (inclusive). Patients were required to be diagnosed with MDD, ≥18 years old, and not diagnosed with bipolar disorder or schizophrenia. HCP specialties included primary care and psychiatry
- Recordings with potential positive insomnia reports were identified via keyword search and confirmed by human review of the full transcript
 - Keywords used to identify potential insomnia symptom reports included: ***Insomnia, Sleep, Asleep, Slept, Sleepless, Restless, Night, Nighttime, Wake, Waking, Woke, Awoke, Awoken***
- Qualitative and quantitative analyses, grounded in the principles of sociolinguistics and conversation analysis were performed; qualitative variables were coded using a mix of inductive and deductive coding approaches

Results

Figure 1: Insomnia and General Sleep-Related Symptom Assessment Across 396 Recordings

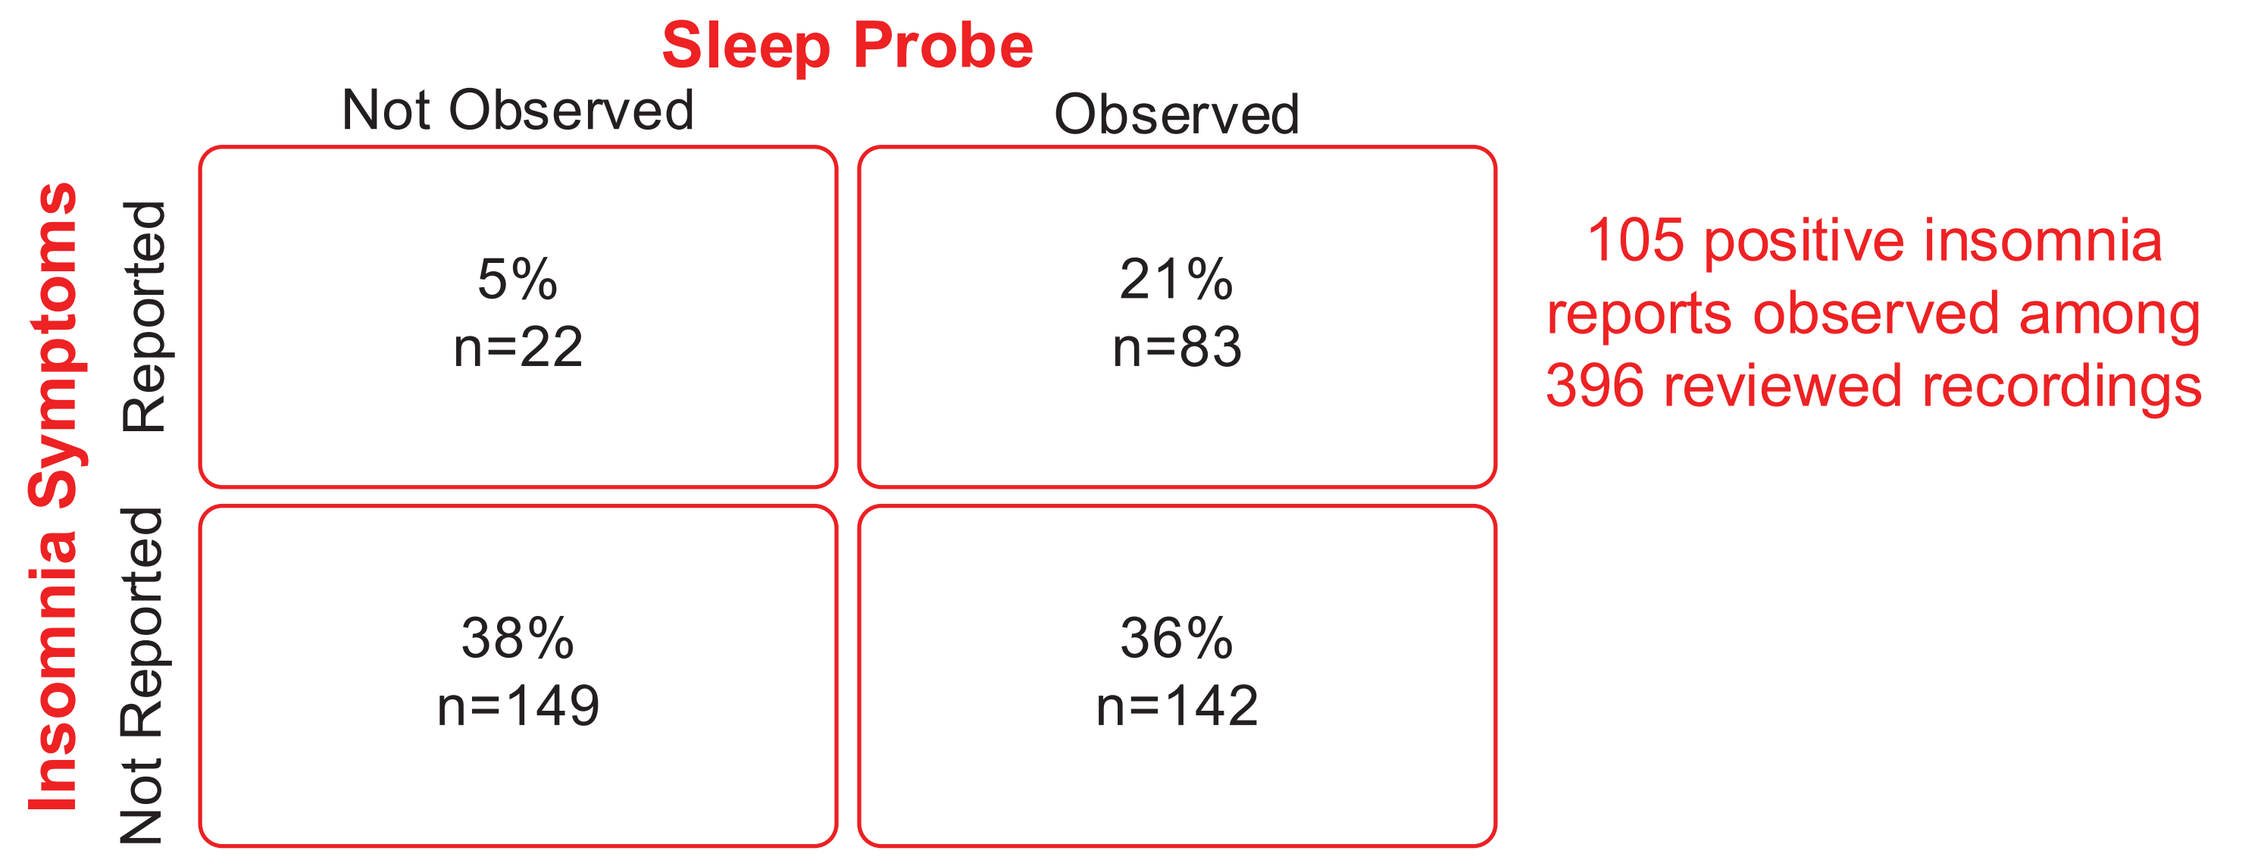
Sleep Probe
Not Observed
Observed
Insomnia Symptoms
Reported
Not Reported
5%
n=22
21%
n=83
38%
n=149
36%
n=142
105 positive insomnia reports observed among 396 reviewed recordings

- Recordings (N=396) were classified along 2 conditions: whether an insomnia or general sleep-related probe was observed during the encounter and whether a positive insomnia symptom report was observed
- Recordings with positive insomnia symptoms (n=105) were carried forward for further analysis
- The analysis included 105 recordings from 25 unique physicians (50 PCP and 55 psychiatrist visits)
- Most visits were in person (n=89/105; 85%), including all PCP visits (50/50), while the 16 telemedicine visits (15%) were conducted exclusively by psychiatrists (n=16/55; 29%)

Figure 2: Time Since Diagnosis of Depression

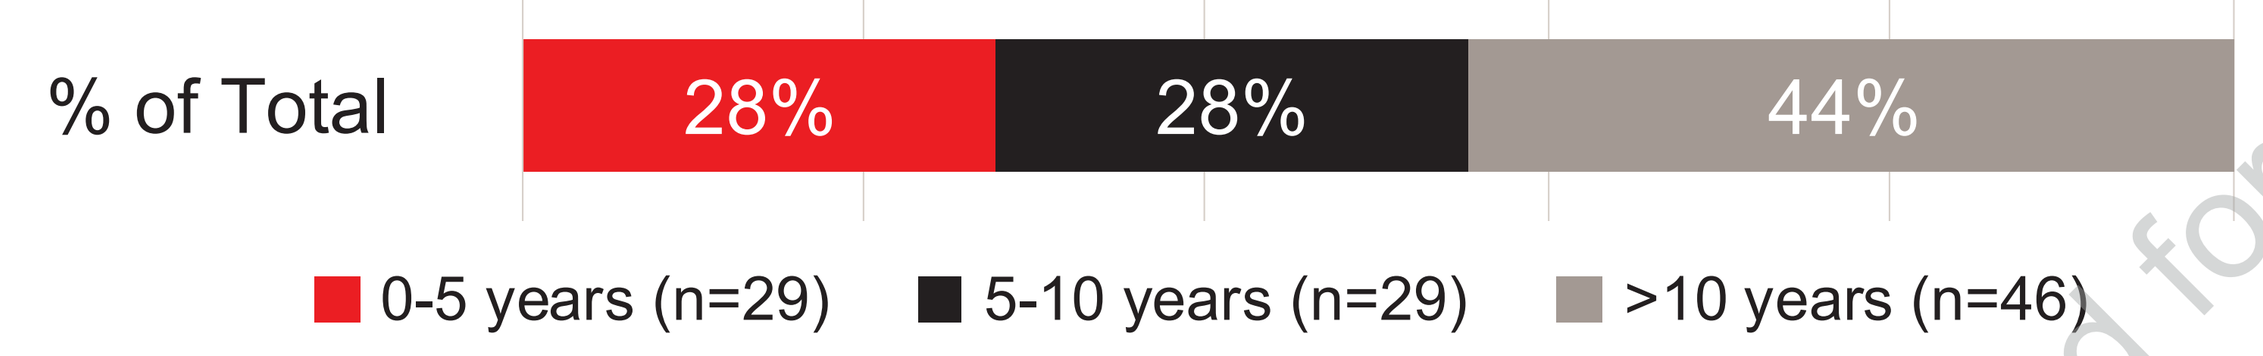
% of Total
28%
28%
44%
0-5 years (n=29)
5-10 years (n=29)
>10 years (n=46)

Figure 3: Who First Brought Up the Topic of Insomnia or General Sleep-Related Symptoms?

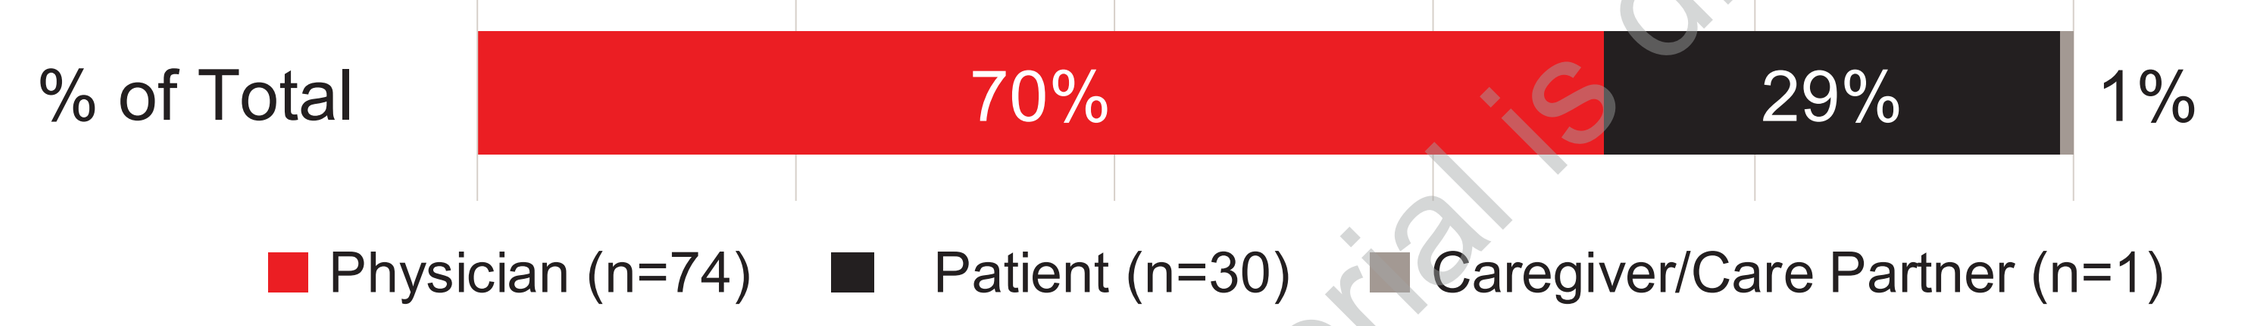
% of Total
70%
29%
1%
Physician (n=74)
Patient (n=30)
Caregiver/Care Partner (n=1)

Figure 4: Recorded Visits Including ≥1 Insomnia or General Sleep-Related Probe (N=105)

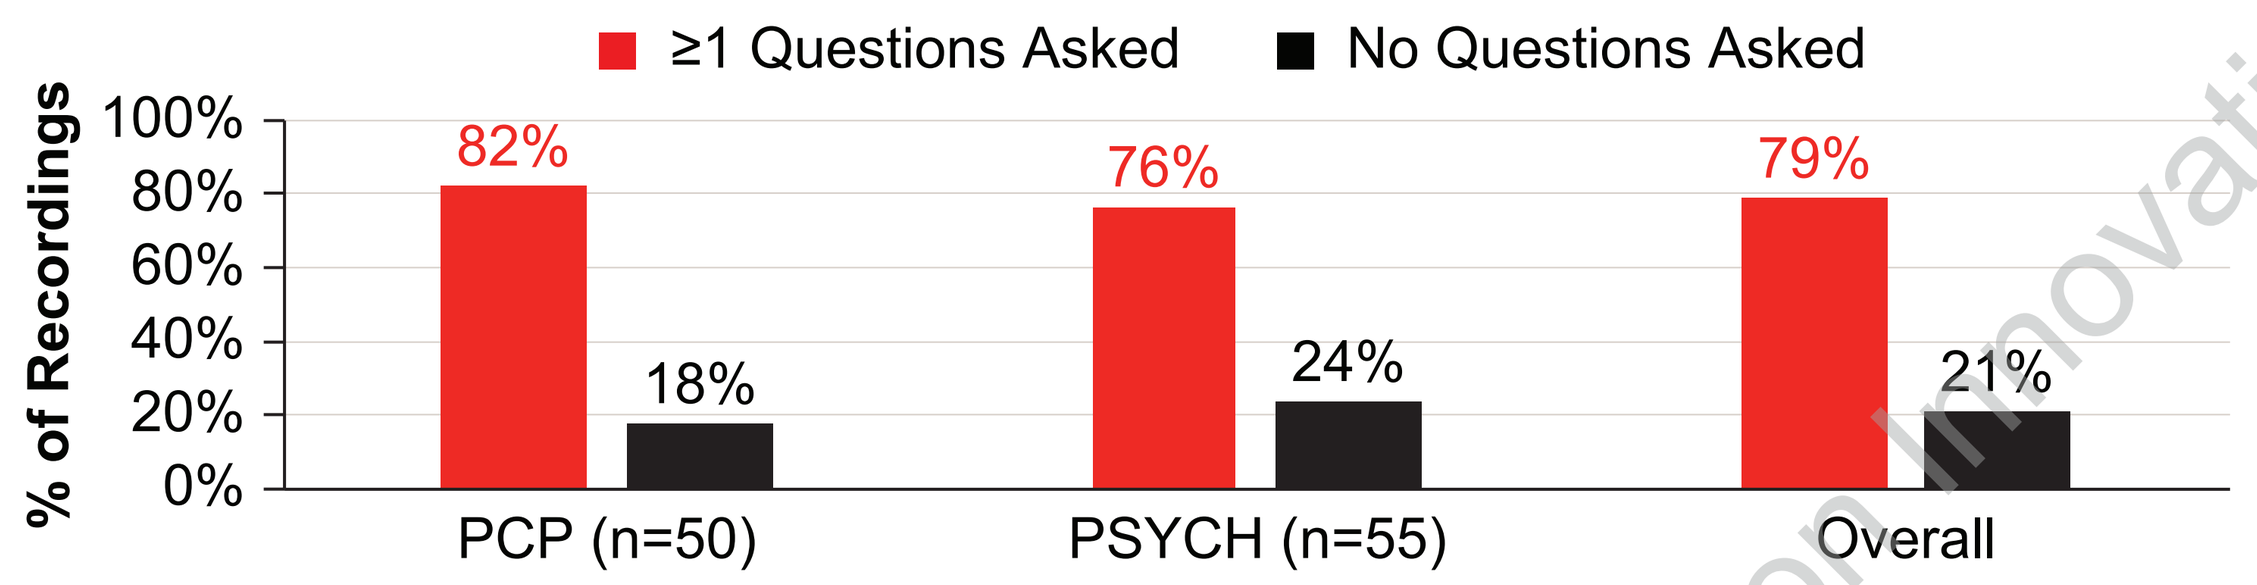
≥1 Questions Asked
No Questions Asked
% of Recordings
100%
80%
60%
40%
20%
0%
82%
18%
76%
24%
79%
21%
PCP (n=50)
PSYCH (n=55)
Overall

- Physicians asked at least 1 insomnia or general sleep-related question in 83 of 105 recordings (79%)
- In <1% of visits (n=1), the physician described any relationship between MDD and insomnia

Figure 5: Types of Probes Used to Assess Insomnia and General Sleep-Related Symptoms

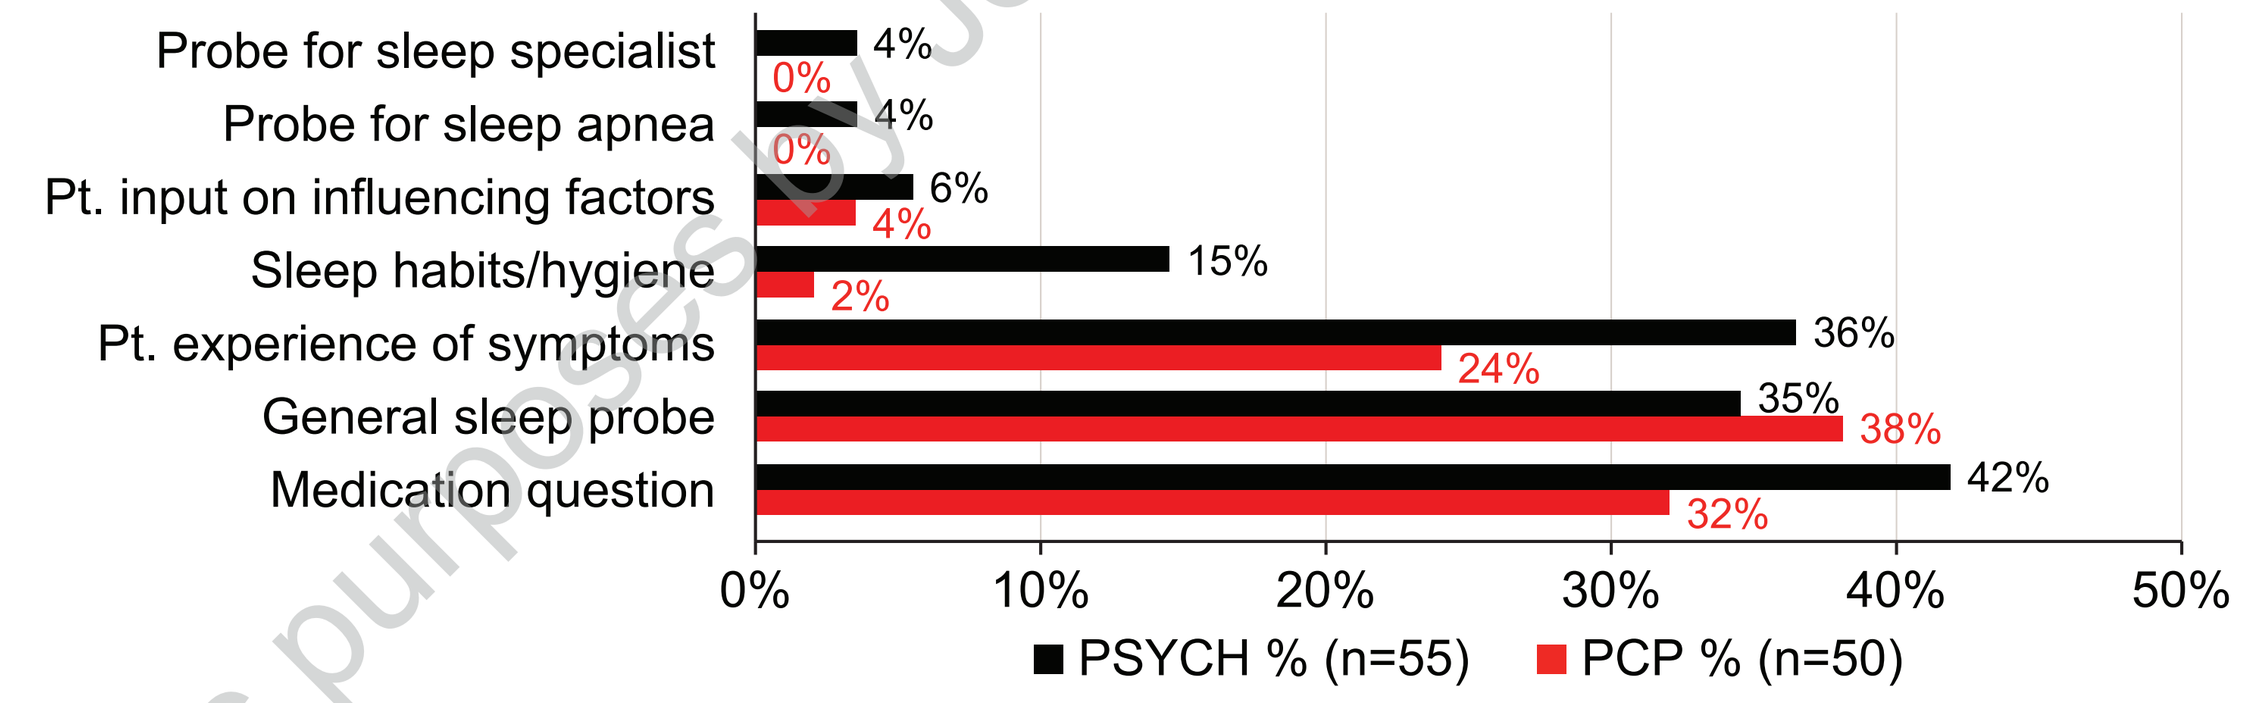
Probe for sleep specialist
4%
0%
Probe for sleep apnea
4%
0%
Pt. input on influencing factors
6%
4%
Sleep habits/hygiene
15%
2%
Pt. experience of symptoms
36%
24%
General sleep probe
35%
38%
Medication question
42%
32%
0%
10%
20%
30%
40%
50%
PSYCH % (n=55)
PCP % (n=50)

- Percentages represent the proportion of recordings containing each identified probe type, stratified by psychiatrists and primary care physicians. A single recording may have included more than one probe type
- The mean number of assessment probes per recording was higher among psychiatrists than primary care physicians (2.3 vs 1.2, respectively)

Table 1: Examples of Questions Used to Assess Insomnia or General Sleep-Related Symptoms

Probe Topic	Verbatims
Medication	*Does the trazodone help you sleep at all?*
General sleep	*Are you sleeping well?*
Pt. experience	*Do you have trouble falling asleep or staying asleep?*
Sleep habits/hygiene	*Do you take some naps during the day?*
Pt. input on influencing factors	*Do you feel like the sleep is because of the moving out or not?*

Figure 6: Daily Life Impact Reported by Patients

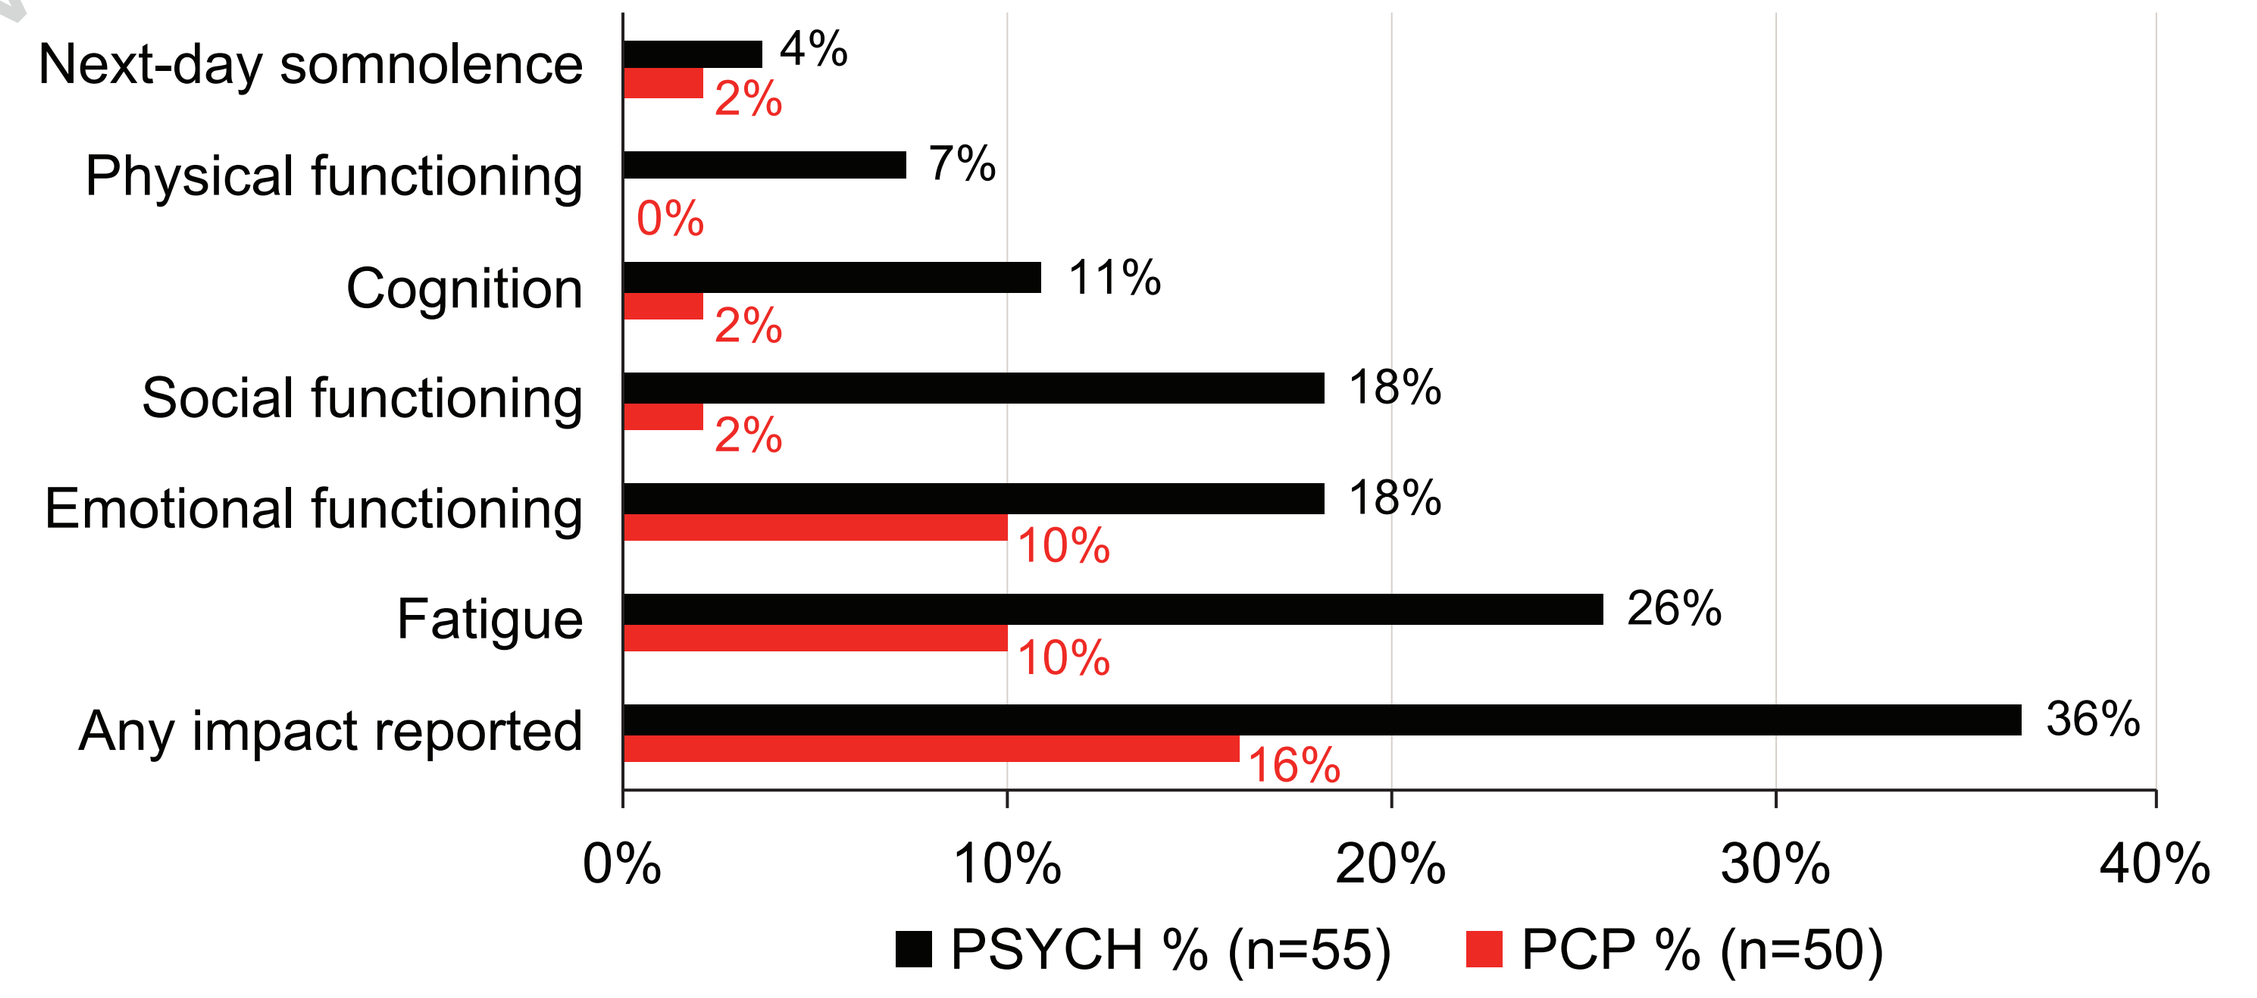
Next-day somnolence
4%
2%
Physical functioning
7%
0%
Cognition
11%
2%
Social functioning
18%
2%
Emotional functioning
18%
10%
Fatigue
26%
10%
Any impact reported
36%
16%
0%
10%
20%
30%
40%
PSYCH % (n=55)
PCP % (n=50)

- Graph shows patient-reported daily life impacts of MDD and/or insomnia symptoms during routine clinical visits

Table 2: Examples of Daily Life Impact Reported by Patients

Impact Type	Verbatims
Fatigue	*I still do feel kinda tired though, even when I do get rest.*
Emotional functioning	*I just cry all the time … at the drop of a hat.*
Social functioning	*I feel … like I literally just want to be left alone. I don't want to talk to nobody. I just want to lock myself in the room and not bother anybody and nobody bothers me.*

Figure 7: Percentage Distribution of Discussed Treatments by Therapeutic Target

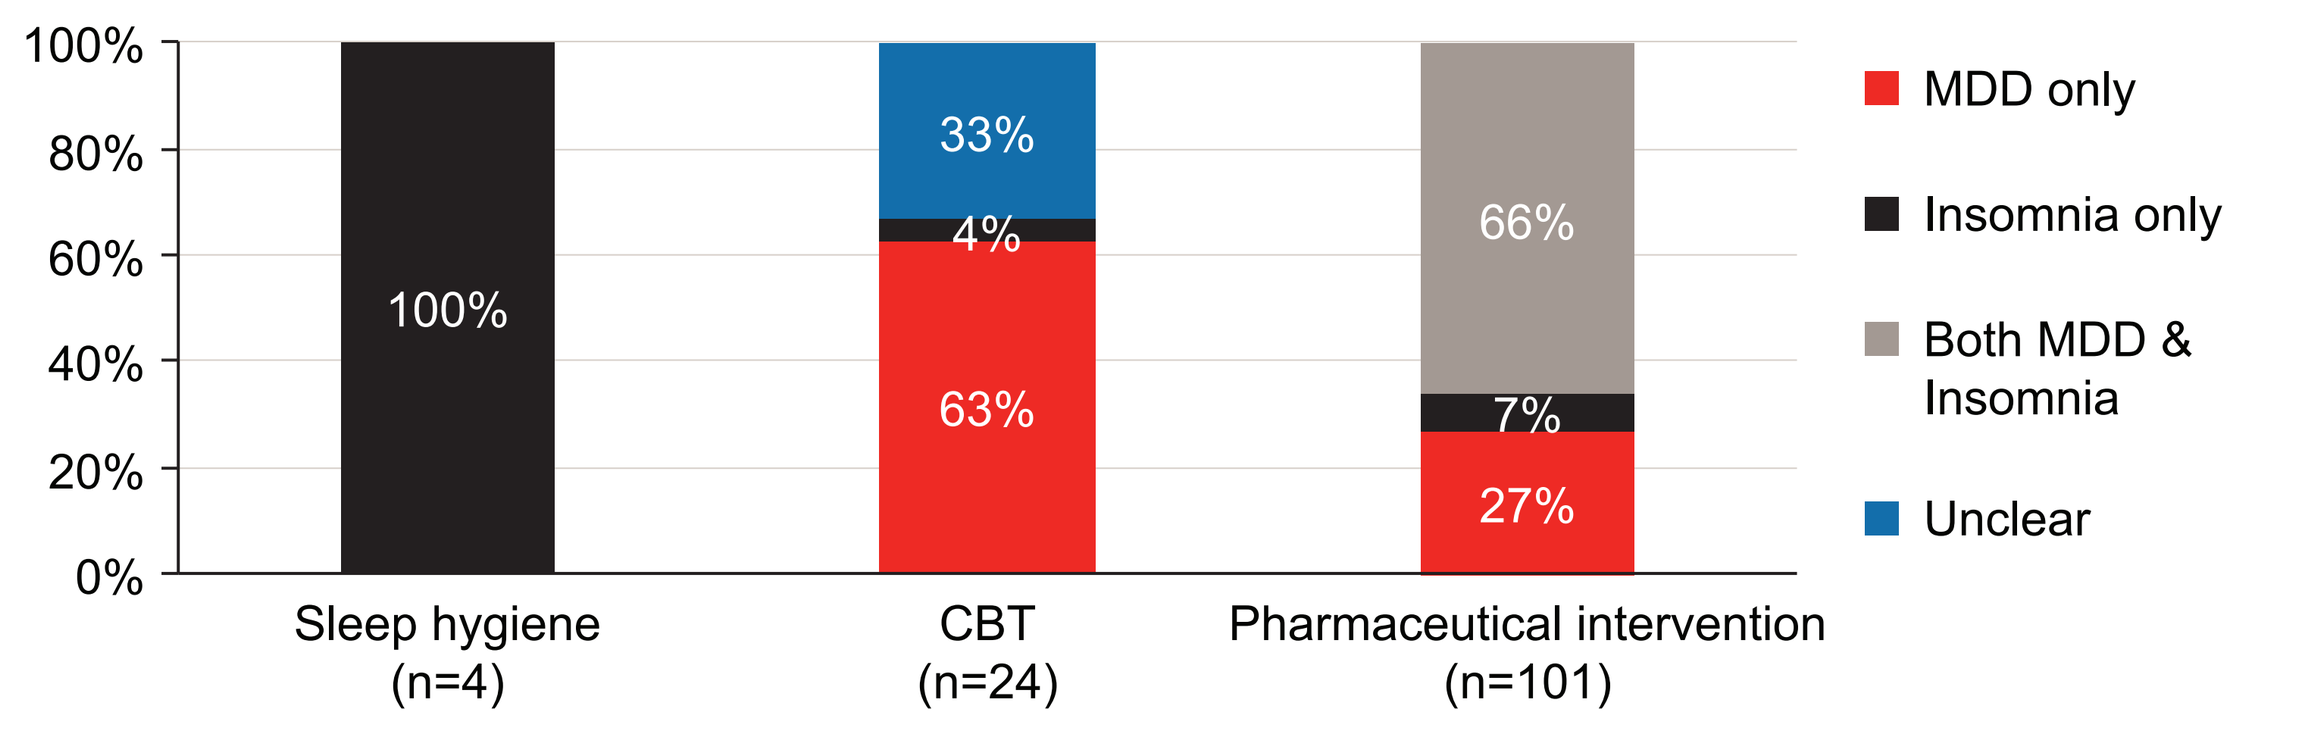
100%
80%
60%
40%
20%
0%
100%
33%
4%
63%
66%
7%
27%
Sleep hygiene
(n=4)
CBT
(n=24)
Pharmaceutical intervention
(n=101)
MDD only
Insomnia only
Both MDD & Insomnia
Unclear

- Only treatment options and targets mentioned by the physician were included in this analysis; patient-only mentions were not counted
- A single recording could include mentions of multiple treatment types
- Treatment was not discussed in 4% (n=4) of the 105 recordings

Key Takeaway

These results underscore opportunities to improve the education, detection, assessment, and management surrounding insomnia symptoms in MDD

Conclusions

- Although insomnia is a core symptom of MDD, an insomnia or general sleep-related probe was observed in only 57% of MDD conversations (N=396), indicating inconsistent assessment of insomnia in routine clinical practice
- Patient-reported daily-life impacts were infrequent and uneven across specialties (36% psychiatry vs 16% PCP)
- Discussion of any link between MDD and insomnia was rare
- Nearly all treatment discussions involved pharmacologic interventions, indicating that medication-based management remains the predominant focus of routine MDD and insomnia symptom treatment

Strengths and Limitations

Strengths

- Directly recorded, in-person visits avoid recall bias inherent to interviews/journaling
- HCP-led consenting/recording minimizes researcher observer effects

Limitations

- Findings should be interpreted as directional, given that the analysis was based on secondary Verilogue market research data from a non-representative sample
- While inductive coding enables data-driven insights, it may pose consistency challenges, and the findings may be limited to conversations of a similar nature

Acknowledgments

The authors acknowledge the contributions of Zachary N. Hebert, MA, and Samantha Creel, PhD, of Verilogue, Philadelphia, PA, USA, for data analysis and development of the original study report. The authors also thank *p*-value communications, Cedar Knolls, NJ, USA, for providing editorial support. This support was funded by Janssen Scientific Affairs, LLC, in accordance with Good Publication Practice Guidelines 2022.

Disclosures

HK, KJ, and HZ are employees of Janssen Scientific Affairs, LLC, and stockholders of Johnson & Johnson, Inc. ZNH and SC are employees of Verilogue, Philadelphia, PA, USA, which received funding from Janssen Scientific Affairs, LLC, to conduct the research and support the development of this abstract.

Novel Pathways in Depression

Scan the QR code

Copies of this poster obtained through Quick Response (QR) Code are for personal use only and may not be reproduced without permission from Psych Congress 2026 or the author of this poster.

Presented at Psych Congress; September 15-19, 2026; New Orleans, LA, USA

Abbreviations:

MDD, major depressive disorder; CBT, cognitive behavioral therapy; HCP, healthcare provider; PCP, primary care physician; PSYCH, psychiatrist; Pt., patient.

References:

1. Major depression. National Institutes of Mental Health (NIMH). 2003. Accessed 16 January 2026. https://www.nimh.nih.gov/health/statistics/major-depression 2. Joshi K, et al. The real-world burden of adults with major depressive disorder with moderate or severe insomnia symptoms in the United States. *J Affect Disord*. 2023;323:698-706. 3. Soehner AM, et al. Prevalence and clinical correlates of co-occurring insomnia and hypersomnia symptoms in depression. *J Affect Disord.* 2014;167:93-97. 4. Zimmerman M, et al. Understanding the severity of depression: which symptoms of depression are the best indicators of depression severity? *Compr Psychiatry.* 2018;87:84-88. 5. Sakurai H, et al. Predicting relapse with individual residual symptoms in major depressive disorder: a reanalysis of the STAR*D data. *Psychopharmacology (Berl).* 2017;234(16):2453-2461. 6. Kale H, et al. Economic and clinical burden of major depressive disorder with insomnia symptoms in commercially and Medicaid-insured adults in the United States: a retrospective matched cohort study. *J Affect Disord*. 2025;372:653-664. 7. Nutt D, et al. Sleep disorders as core symptoms of depression. *Dialogues Clin Neurosci.* 2008;10(3):329-336. 8. Peterson MJ, Benca RM. Sleep in mood disorders. *Psychiatr Clin North Am.* 2006;29(4):1009-1032; abstract ix.