97

Rapid and Sustained Remission With Esketamine Nasal Spray in Adult Patients With Treatment-Resistant Depression: Overview of Remission Rates From Six Clinical Trials

Rakesh Jain[1], Dong-Jing Fu[2], Ibrahim Turkoz[2], Mai Himedan[2], Lisa Lim[2], Oliver Lopena[2], Ronaldo Naranjo Jr.[2], Lilianna Ly[2], Alexis Davis[2], Allen Wu[2], Kristian Dambrino[3]

[1]Texas Tech University School of Medicine—Permian Basin, Midland, TX; [2]Johnson & Johnson, Titusville, NJ; [3]Belmont University College of Nursing, Nashville, TN

This material is distributed for scientific purposes by Johnson & Johnson Innovative Medicine and is not for promotional use.

Introduction

- Attaining remission in patients with major depressive disorder (MDD) reduces risk for relapse, improves functioning, and reduces morbidity and mortality[1]
- However, achieving remission in patients with treatment-resistant depression (TRD), defined as inadequate response to ≥2 antidepressants, is challenging owing to prior treatment failures, chronic illness, comorbidities, and persistent residual symptoms[1,2]
 - In step 3 of the STAR*D trial (consistent with the definition of TRD), only 13.7% of patients attained remission, as defined by Quick Inventory of Depressive Symptomatology Self-Report (QIDS-SR) total score ≤5[3]
- Esketamine nasal spray (ESK) is approved for the treatment of:
 - TRD in adults, as monotherapy or in conjunction with an oral antidepressant (OAD)[4]
 - Depressive symptoms in adults with MDD with acute suicidal ideation or behavior in conjunction with an OAD[4]
- Across TRD clinical trials, treatment with ESK is associated with rapid and sustained remission[5-11]; however, a comprehensive overview of remission rates among patients who received ESK is currently lacking
- The objective of this analysis was to summarize remission rates achieved with ESK, as defined by Montgomery-Åsberg Depression Rating Scale (MADRS) total score ≤10 and ≤12, across 6 TRD clinical trials among patients who received ESK consistent with US prescribing information[4]

Methods

Study designs

- Data were analyzed from 6 ESK TRD trials (**Table 1**)
 - Two 4-week placebo-controlled ESK trials
 - TRD4005 (phase 4, double-blind, randomized, monotherapy trial of ESK 56 mg or 84 mg vs placebo nasal spray [PBO])[10]
 - TRANSFORM-2 (phase 3, double-blind, randomized trial of ESK 56 mg or 84 mg + OAD vs PBO + OAD)
 - One active-controlled trial, ESCAPE-TRD (phase 3b, open-label, single-blind trial of ESK + selective serotonin reuptake inhibitor [SSRI]/serotonin-norepinephrine reuptake inhibitor [SNRI] vs quetiapine extended-release [QUE XR] + SSRI/SNRI)
 - Three long-term open-label trials
 - ESCAPE-LTE (phase 4, single-arm, open-label extension trial of ESK + SSRI/SNRI)
 - SUSTAIN-2 (phase 3, open-label trial of ESK + OAD)
 - SUSTAIN-3 (phase 3, open-label, long-term extension trial of ESK + OAD)

Assessments

- Analyses included patients that were administered ESK consistent with US prescribing information[4]
- Baseline demographics and disease characteristics of patients from each study are summarized descriptively
- Remission was defined as MADRS total score ≤10 and ≤12
- Safety data are summarized descriptively

Table 1: Overview of ESK TRD study designs

Trial type	Study	Design	Treatment	Duration	Primary endpoint
PBO-controlled trials	TRD4005 (NCT04599855)	Phase 4, DB, randomized, PBO control	• ESK 56 or 84 mg • PBO	DB: 28 days OL: 12 weeks	Change in MADRS total score from BL to day 28
	TRANSFORM-2 (NCT02418585)	Phase 3, DB randomized, PBO control	• ESK 56 or 84 mg + OAD • PBO + OAD	28 days	Change from BL in MADRS at day 28
Active-controlled trial	ESCAPE-TRD (NCT04338321)	Phase 3b, OL, SB, active control	• ESK + SSRI/SNRI • QUE XR + SSRI/SNRI	32 wks	Remission (MADRS score ≤10) at wk 8
Open-label trials	ESCAPE-LTE (NCT04829318)	Phase 4, single-arm OL extension	• ESK + SSRI/SNRI	136 wks	Safety
	SUSTAIN-2 (NCT02497287)	Phase 3, OL	• ESK + OAD	IND: 4 wks OP/M: 48 wks	Safety
	SUSTAIN-3 (NCT02782104)	Phase 3, OL, LTE	• ESK + OAD	IND: 4 wks OP/M: variable	Safety

BL, baseline; DB, double blind; ESK, esketamine nasal spray; IND, induction phase; LTE, long-term extension; MADRS, Montgomery-Åsberg Depression Rating Scale; OAD, oral antidepressant; OL, open label; OP/M; optimization/maintenance phase; PBO, placebo nasal spray; QUE XR, quetiapine extended-release; SB, single blind; SSRI, selective serotonin reuptake inhibitor; SNRI, serotonin-norepinephrine reuptake inhibitor; TRD, treatment-resistant depression; wk, week.

Results

Baseline characteristics

- Baseline demographics and disease characteristics of patients are shown in **Table 2**
- Mean age ranged from 41.8 to 47.3 years, and most patients were female (57.8%-69.4%)
- Patients had TRD and moderate to severe depression in their current episode, with baseline MADRS total scores ranging from 29.1 to 37.6

Table 2: Baseline demographics and disease characteristics

	Placebo-controlled trials					Active-controlled trial		Open-label trials		
	TRD4005			TRANSFORM-2		ESCAPE-TRD		ESCAPE-LTE	SUSTAIN-2	SUSTAIN-3
	ESK 56 mg	ESK 84 mg	PBO	ESK+OAD	PBO+OAD	ESK+SSRI/SNRI	QUE XR +SSRI/SNRI	ESK+SSRI/SNRI	ESK+OAD	ESK+OAD
N	76	85	180	114	109	314	320	173	473	1021[a]
Age, mean (SD)	43.4 (11.94)	41.8 (12.38)	43.1 (12.46)	44.9 (12.58)	46.4 (11.14)	42.8 (12.56)	44.5 (12.40)	43.3 (12.18)	46.6 (10.98)	47.3 (10.71)
Age at MDD Dx, mean (SD)	23.6 (9.93)	24.4 (9.80)	25.5 (11.22)	32.1 (12.53)	35.3 (13.04)	33.0 (11.61)	34.4 (11.67)	34.1 (11.26)	33.1 (11.79)	32.8 (11.9)
Female, n (%)	49 (64.5)	53 (62.4)	107 (59.4)	75 (65.8)	63 (57.8)	211 (67.2)	207 (64.7)	120 (69.4)	300 (63.4)	678 (66.4)
BL MADRS total score, mean (SD)	37.6 (5.30)	37.0 (4.39)	37.6 (4.90)	37.0 (5.69)	37.3 (5.66)	31.6 (6.03)	31.1 (5.94)	31.6 (5.63)	31.2 (4.77)	29.1 (7.94)[b]
BL PHQ-9 total score, mean (SD)	20.9 (3.36)	20.1 (3.64)	19.9 (4.00)	20.2 (3.63)	20.4 (3.74)	17.9 (4.26)	17.4 (4.56)	17.4 (3.97)	17.8 (4.61)	15.4 (5.56)[c]

BL, baseline; Dx, diagnosis; ESK, esketamine nasal spray; MADRS, Montgomery-Åsberg Depression Rating Scale; MDD, major depressive disorder; OAD, oral antidepressant; PBO, placebo nasal spray; PHQ-9, 9-item Patient Health Questionnaire; QUE XR, quetiapine extended-release; SNRI, serotonin-norepinephrine reuptake inhibitor; SSRI, selective serotonin reuptake inhibitor.
[a]All enrolled analysis set.
[b]Data from Zajecka et al.[12]
[c]Baseline induction score (N = 439).

Remission rates based on MADRS total score ≤10

Placebo-controlled ESK trials

- Remission was assessed at week 4 in both placebo-controlled ESK trials (**Figure 1**)
 - In TRD4005, remission rates were 6.5%, 13.9%, and 21.5%, with PBO, ESK 56 mg, and ESK 84 mg, respectively
 - In TRANSFORM-2, remission rates were 24.0% in the PBO + OAD group vs 42.6% in the ESK + OAD group
- In TRD4005, remission rates at week 16 endpoint were 32.7% with PBO/ESK and 32.2% with ESK/ESK (**Figure 1**)

Active-controlled ESK trial

- In the ESCAPE-TRD trial, remission rates were assessed at week 8 and week 32 (**Figure 1**)
- At week 8, remission was attained in 22.9% of patients who received QUE XR + SSRI/SNRI and 29.4% of patients who received ESK + SSRI/SNRI
- At week 32, remission rates were 54.2% vs 64.6%, respectively

Open-label ESK trials

- In open-label ESK trials, remission was assessed at week 48, week 136, and SUSTAIN-3 OP/M endpoint (~5.5 years) (**Figure 1**)
- In SUSTAIN-2, 49.3% of patients receiving ESK + OAD attained remission at week 48 (optimization and maintenance phase [OP/M] endpoint)
- In the long-term extension of ESCAPE, 78.2% of patients receiving ESK + SSRI/SNRI attained remission at week 136
- In SUSTAIN-3, 43.2% of patients receiving ESK + OAD attained remission at OP/M endpoint

Figure 1: Remission rates (MADRS total score ≤10)

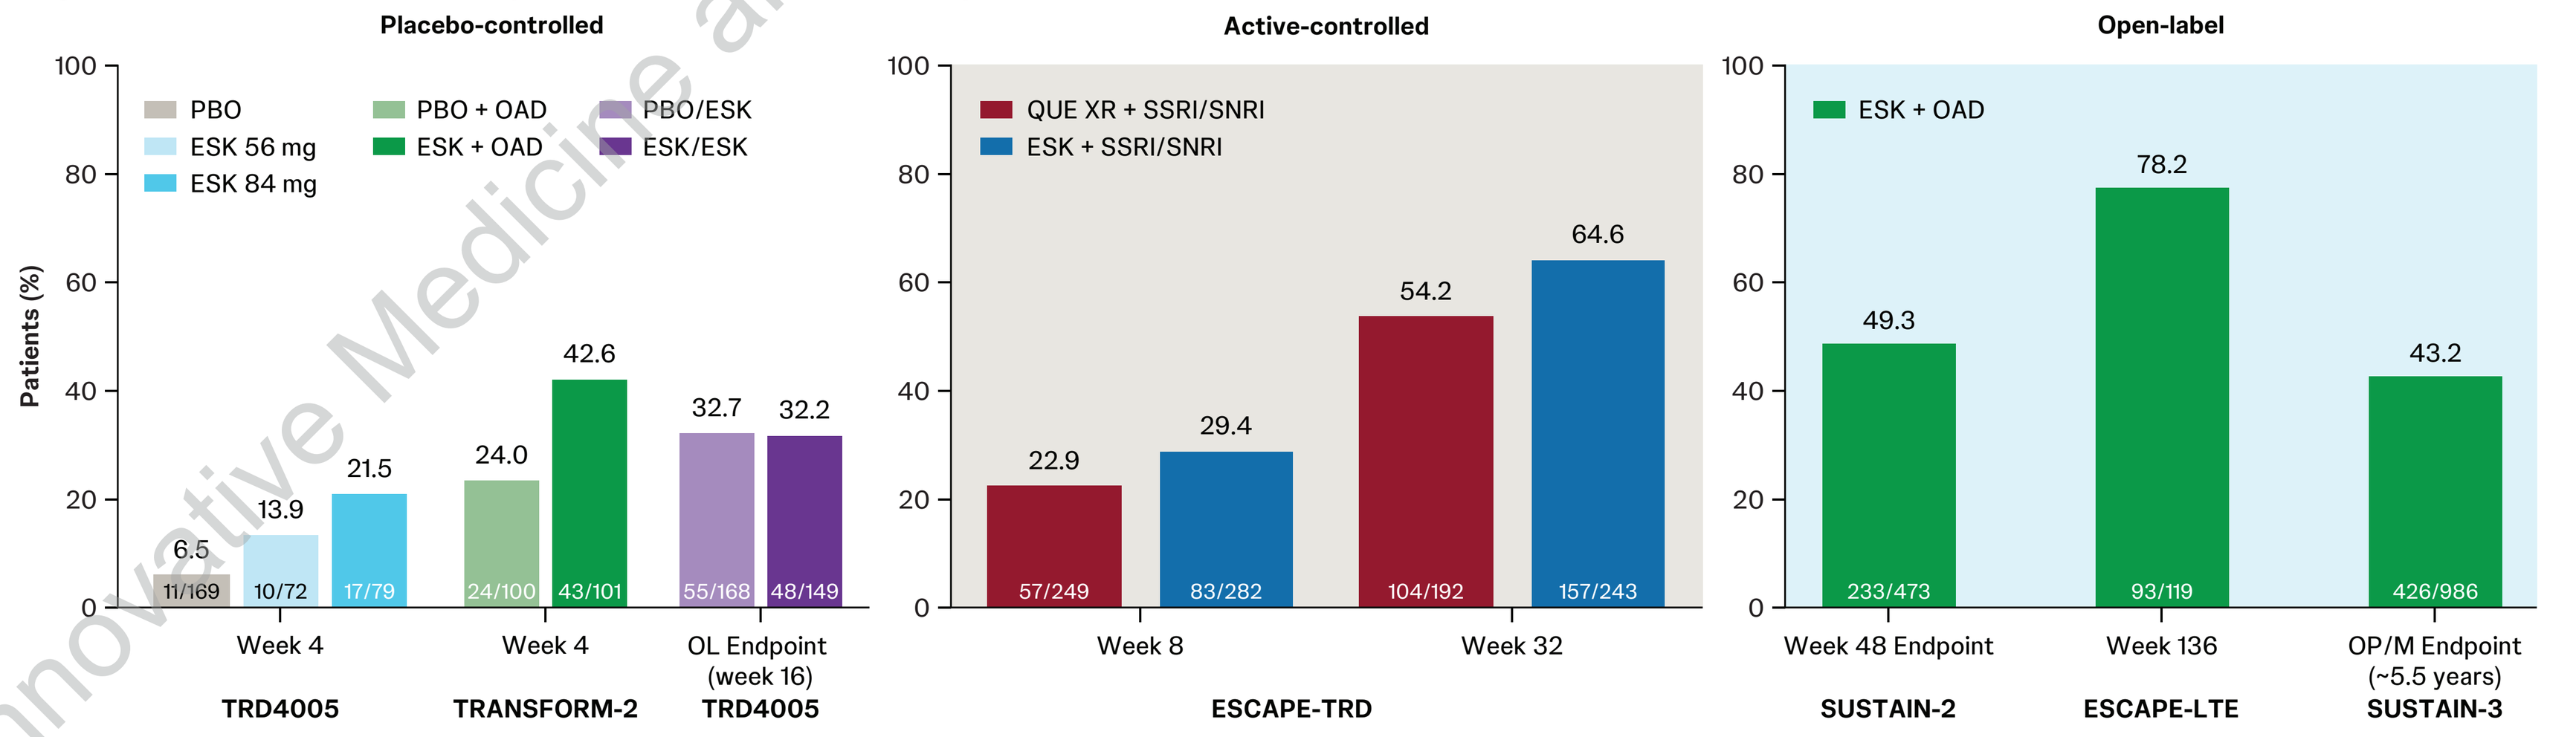
Placebo-controlled
Active-controlled
Open-label
Patients (%)
PBO
ESK 56 mg
ESK 84 mg
PBO + OAD
ESK + OAD
PBO/ESK
ESK/ESK
6.5
13.9
21.5
24.0
42.6
32.7
32.2
11/169
10/72
17/79
24/100
43/101
55/168
48/149
Week 4
Week 4
OL Endpoint (week 16)
TRD4005
TRANSFORM-2
TRD4005
QUE XR + SSRI/SNRI
ESK + SSRI/SNRI
22.9
29.4
54.2
64.6
57/249
83/282
104/192
157/243
Week 8
Week 32
ESCAPE-TRD
ESK + OAD
49.3
78.2
43.2
233/473
93/119
426/986
Week 48 Endpoint
Week 136
OP/M Endpoint (~5.5 years)
SUSTAIN-2
ESCAPE-LTE
SUSTAIN-3

ESK, esketamine nasal spray; ESK/ESK: patients who received ESK after 4 weeks of ESK (56 mg or 84 mg); OAD, oral antidepressant; OP/M; optimization/maintenance phase; PBO, placebo nasal spray; PBO/ESK, patients who received treatment with ESK after 4 weeks of PBO; QUE XR, quetiapine extended-release; SNRI, serotonin-norepinephrine reuptake inhibitor; SSRI, selective serotonin reuptake inhibitor; TRD, treatment-resistant depression.

Remission rates based on MADRS total score ≤12

Placebo-controlled ESK trials

- In TRD4005, remission rates were
 - 7.7%, 15.3%, and 22.8%, with PBO, ESK 56 mg, and ESK 84 mg at week 4, respectively
 - 36.9% and 35.6% with PBO/ESK and ESK/ESK at week 16, respectively
- In TRANSFORM-2, remission rates were 31.0% (PBO + OAD) vs 52.5% (ESK + OAD) at week 4

Active-controlled ESK trial

- In the ESCAPE-TRD trial, remission rates were 29.7% (QUE XR + SSRI/SNRI) vs 42.6% (ESK + SSRI/SNRI) at week 8 and 67.7% vs 74.5%, respectively, at week 32

Open-label ESK trials

- In SUSTAIN-2, 56.2% of patients receiving ESK + OAD attained remission at week 48 (OP/M endpoint)
- In the long-term extension of ESCAPE, 86.6% of patients receiving ESK + OAD attained remission at week 136
- In SUSTAIN-3, 49.6% of patients receiving ESK + OAD attained remission at OP/M endpoint (~5.5 years)

Safety

- The incidence of treatment emergent adverse events (AEs), serious adverse events (SAEs), and most common AEs for each study are shown in **Table 3**
- Rates of dissociation, nausea, dizziness and headache are consistent with the known safety profile for ESK

Table 3: Safety summary

	Placebo-controlled trials				Active-controlled trial		Open-label trials		
	TRD4005[a]		TRANSFORM-2		ESCAPE-TRD		ESCAPE-LTE	SUSTAIN-2	SUSTAIN-3
	ESK	PBO	ESK+OAD	PBO+OAD	ESK+SSRI/SNRI	QUE XR +OAD	ESK+SSRI/SNRI	ESK+OAD	ESK+OAD
N	161	180	114	109	314	320	173	473	1022
≥1 TEAEs, n (%)	117 (72.7)	86 (47.8)	96 (84.2)	66 (60.6)	289 (92.0)	248 (77.5)	151 (87.3)	440 (93.0)	968 (94.7)
≥1 SAE, n (%)	2 (1.2)	2 (1.1)	1 (0.9)	1 (0.9)	19 (6.1)	15 (4.7)	11 (6.4)	34 (7.2)	184 (18.0)
Most common TEAEs (≥10%)									
Dissociation	3 (27.3)	0 (0)	14 (12.3)	2 (1.8)	89 (28.3)	2 (0.6)	19 (11.0)	142 (30.0)	270 (26.4)
Nausea	4 (36.4)	0 (0)	30 (26.3)	7 (6.4)	94 (29.9)	10 (3.1)	36 (20.8)	142 (30.0)	352 (34.4)
Dizziness	3 (27.3)	1 (33.3)	23 (20.2)	5 (4.6)	148 (47.1)	25 (7.8)	47 (27.2)	170 (35.9)	355 (34.7)
Headache	2 (18.2)	0 (0)	21 (18.4)	19 (17.4)	80 (25.5)	41 (12.8)	76 (43.9)	155 (32.8)	383 (37.5)

ESK, esketamine nasal spray; OAD, oral antidepressant; PBO, placebo nasal spray; QUE XR, quetiapine extended-release; SAE, serious adverse event; SNRI, serotonin-norepinephrine reuptake inhibitor; SSRI, selective serotonin reuptake inhibitor; TEAE, treatment-emergent adverse event.
[a]Safety data are from CFL analysis set from double-blind phase.

Key Takeaways

- Achieving remission in patients diagnosed with MDD or TRD is a critical goal in clinical practice
- Rapid and sustained remission, defined as MADRS total score ≤10 and ≤12, was observed across 6 TRD studies with ESK treatment, administered as monotherapy or in combination with an OAD, supporting remission as an attainable treatment goal for patients with TRD

Limitations

- This was not a formal meta-analysis, and no pooled effect estimates or statistical comparisons were performed
- Differences in patient populations and trial designs limit direct comparability

Conclusions

- Remission rates across 6 studies of esketamine nasal spray for TRD demonstrate rapid and sustained remission when administered as monotherapy and in combination with an OAD
 - As monotherapy, remission rates (MADRS total score ≤10) ranged from 13.9% to 21.5% at week 4 and 32.2% at week 16
 - As combination therapy (esketamine nasal spray + OAD), remission rates (MADRS total score ≤10) ranged from 29.4% to 78.2% over treatment durations up to ~5.5 years
- Attainment of remission was consistently observed across esketamine nasal spray TRD studies with varying designs
 - Inclusion of trials with and without a concomitant OAD reflects real-world treatment scenarios
 - Both short-term (induction) and longer-term (maintenance) data allowed evaluation of both initial response and sustained remission

Acknowledgments

The authors thank Lynn Brown, PhD, CMPP (ApotheCom, Yardley, PA), for editorial and writing assistance, which was funded by Johnson & Johnson.

Disclosures

Dr Jain: RD reports acting as a consultant for AbbVie, Acadia, Adamas, Alfasigma, Alkermes, Almatica, Axsome, Biogen, Boehringer Ingelheim, Cingulate Therapeutics, Corium, Eisai, Evidera, Impel, Janssen, Lilly, Lundbeck, Merck, Neos Therapeutics, Neurocrine Biosciences, Osmotica, Otsuka, Pamlab, Pfizer, Sage Therapeutics, Shire, Sumitomo, Sunovion, Supernus, Takeda, Teva, Transcend Therapeutics, and Viatris; receiving research support from AbbVie, Lilly, Lundbeck, Otsuka, Pfizer, Shire, and Takeda; being a member of speakers bureaus for AbbVie, Alkermes, Almatica, Axsome, Corium, Eisai, Intra-Cellular Therapies, Ironshore Pharmaceuticals, Janssen, Lilly, Lundbeck, Merck, Neos Therapeutics, Otsuka, Pamlab, Pfizer, Shire, Sumitomo, Sunovion, Takeda, Tris Pharmaceuticals, and Viatris; and participating on advisory boards for Adamas, Alkermes, Corium, Eisai, Janssen, Lilly, Lundbeck, Merck, Neos Therapeutics, Neurocrine Biosciences, Otsuka, Pamlab, Sage Therapeutics, Shire, Sunovion, Supernus, Teva, and Usona. D-JF, IT, MH, L Lim, OL, RN, L Ly, AD, and AW are employees of Johnson & Johnson, Titusville, NJ. KD reports participating on advisory boards for Alkermes, Axsome Therapeutics Inc., Johnson & Johnson, and Neurocrine Biosciences; acting as a consultant for AbbVie Inc., Axsome Therapeutics Inc., Johnson & Johnson, and Neurocrine Biosciences; being a member of speakers bureaus for Alkermes, Axsome Therapeutics Inc., Bristol Myers Squibb, Intra-Cellular Therapies (Johnson & Johnson), Luye Pharma Group, and Neurocrine Biosciences.

Previously presented at Psych Congress Elevate; June 3-6, 2026; Las Vegas, NV, USA

Novel Pathways in Depression

Scan the QR code

The QR code is intended to provide scientific information for individual reference, and the information should not be altered or reproduced in any way.

This study was funded by Johnson & Johnson.

Presented at Psych Congress; September 15-19, 2026; New Orleans, Louisiana, USA

References

1. Oliveira-Maia AJ et al. *Adv Ther*. 2024;41(1):34-64. 2. Judd L et al. *Am J Psychiatry*. 2000;157(9):1501-1504. 3. Rush AJ et al. *Am J Psychiatry*. 2006;163(11):1905-1917. 4. SPRAVATO® (esketamine) nasal spray, CIII. Prescribing information. Janssen Pharmaceuticals, Inc.; 2026. 5. Popova V et al. *Am J Psychiatry*. 2019;176(6):428-438. 6. Ochs-Ross R et al. *Am J Geriatr Psychiatry*. 2020;28(2):121-141. 7. Zaki N et al. *Int J Neuropsychopharmacol*. 2025;28(6): pyaf027. 8. Mattingly G et al. Presented at: Psych Congress Elevate; June 1-4, 2023; Las Vegas, NV. 9. Himedan M et al. Presented at: Neuroscience Education Institute (NEI) Fall Congress; November 6-9, 2025; Colorado Springs, CO. 10. Janik A et al. *JAMA Psychiatry*. 2025;82(9):877-887. 11. Reif A et al. *N Engl J Med*. 2023;389(14):1298-1309. 12. Zajecka J et al. Presented at Psych Congress; September 6–10, 2023; Nashville, TN.