84

Patient and Healthcare Practitioner Treatment Preferences for Treatment-Resistant Depression

Josh Hamilton[1], Hui Lu[2], Laura E Panattoni[3], Anup Das[2], Jennifer A Whitty[2], Brahim Bookhart[4], Dejan Milentijevic[4], Rakesh Amin[5-7]

[1]Washington State University, Spokane, WA, USA; [2]PPD Evidera Patient-Centered Research, Thermo Fisher Scientific, London, UK; [3]PPD Evidera Patient-Centered Research, Thermo Fisher Scientific, Seattle, WA, USA; [4]Johnson & Johnson, Titusville, NJ, USA; [5]Pure Psychiatry Group of Michigan and Ohio, USA, [6]Vital Health Management, Detroit, Michigan, USA, [7]Unity Psychiatric Care, Columbia, TN and Clarksville, TN, USA

Introduction

- Treatment-resistant depression (TRD) is commonly characterized by an inadequate response to at least 2 antidepressants administered at an appropriate dose and duration[1]
- Its management is increasingly complex because available and emerging pharmacologic and brain stimulation treatments differ in efficacy, safety, administration, regulatory status, and the maturity of long-term evidence[2]
- Few studies have compared patient and healthcare professional (HCP) treatment preferences using the same preference-elicitation framework[3-5]
- Understanding how adults with TRD and HCPs value these characteristics can support patient-centered treatment selection, benefit-risk communication, and shared decision-making

Objective

- To quantify and compare treatment preferences and benefit-risk trade-offs among adults with TRD and HCPs managing TRD in the United States

Methods

- A cross-sectional, web-based survey including a discrete choice experiment (DCE) was conducted in the United States[6-8]
- The preference instrument was developed using an evidence review, steering committee input, and cognitive pilot interviews with 15 adults with TRD and 10 HCPs
- The main survey included adults, recruited through a patient panel, with confirmed major depressive disorder and a self-reported history of using at least 2 antidepressants for ≥6 weeks each. The study also included HCPs recruited through an HCP panel, comprising psychiatrists, nurse practitioners, or psychiatric physician associates with ≥3 years of experience managing depression, including TRD, in the US
- Patients and HCPs completed 10 experimental choice tasks comparing 2 hypothetical treatments. Treatment profiles varied across their administration mode and 7 treatment "attributes": US Food and Drug Administration (FDA) approval and long-term safety evidence, onset time, chance of response at 8 weeks and 1 year, and risks of dissociation, sexual dysfunction, and weight gain (see **Figure 1**)
- Mixed logit models estimated preference weights, which were used to calculate the following outputs
 - Relative attribute importance (RAI), which quantifies the percentage contribution of a specified improvement in administration mode and each attribute included in the DCE to treatment decisions. RAI is measured on a scale in which the contributions of all attribute total 100%
 - The minimum additional increase in chance of response at 8 weeks required to accept an increase in a treatment-related risk, a less-preferred administration mode, or a treatment without FDA approval

Figure 1: Example patient DCE choice task

Imagine that you are in a medical appointment to discuss your treatment for depression and your doctor presents two treatment options for you. Please see the example below.

Please review each treatment and indicate which treatment you would select if these were the only two treatments available to you. Please assume the treatments are identical except for the listed features.	Oral Tablets	Nasal Spray
FDA approval and long-term safety data	FDA approved and no long-term safety data	FDA approved with long-term safety data
Onset time (time until some improvement)	3 weeks Within 3 weeks	1 day Within 1 day
Chance of a substantial improvement within 8 weeks	20 out of 100 (20%)	70 out of 100 (70%)
Chance of a substantial improvement at 1 year	5 out of 100 (5%)	35 out of 100 (35%)
Risk of dissociation	0 out of 100 (0%)	40 out of 100 (40%)
Risk of sexual dysfunction	40 out of 100 (40%)	0 out of 100 (0%)
Risk of weight gain	30 out of 100 (30%)	0 out of 100 (0%)
Please choose your preferred option	☐	☐

- Preference heterogeneity was explored using interacted mixed logit models for prespecified subgroups. Prespecified patient subgroup analyses were conducted by age, gender identity, time since TRD diagnosis, previous experience with intranasal esketamine, intravenous ketamine, or transcranial magnetic stimulation (TMS), and number of previous treatments received. Prespecified HCP subgroup analyses were conducted by professional role, gender identity, experience managing patients with TRD, and practice setting

Results

Participant characteristics

- The analysis included 200 adults with TRD and 151 HCPs who completed the survey
- Patients had a mean age of 40.0 years and were predominantly (78.5%) women. Most (78.5%) had been diagnosed with depression for more than 5 years and were currently receiving antidepressants and medications prescribed for antidepressant effect (87.5%)
- HCPs included psychiatrists (58.9%), nurse practitioners (19.9%), and psychiatric physician associates (22.5%), with nearly all reporting more than 5 years of experience managing TRD

Key drivers of treatment preference

- FDA approval with long-term safety evidence, together with chance of response at 1 year and chance of response at 8 weeks, were the most influential treatment characteristics for both patients and HCPs (**Figure 2**, conditional on the specified improvement shown in brackets)
- Patients placed significantly greater importance than HCPs on avoiding dissociation (12.4% vs 8.4%; $P < 0.05$) and weight gain (12.2% vs 7.1%; $P < 0.05$)
- Avoiding intravenous administration and a faster onset time mattered but had the lowest relative importance for both groups

Figure 2: RAI: comparison between patients and HCPs

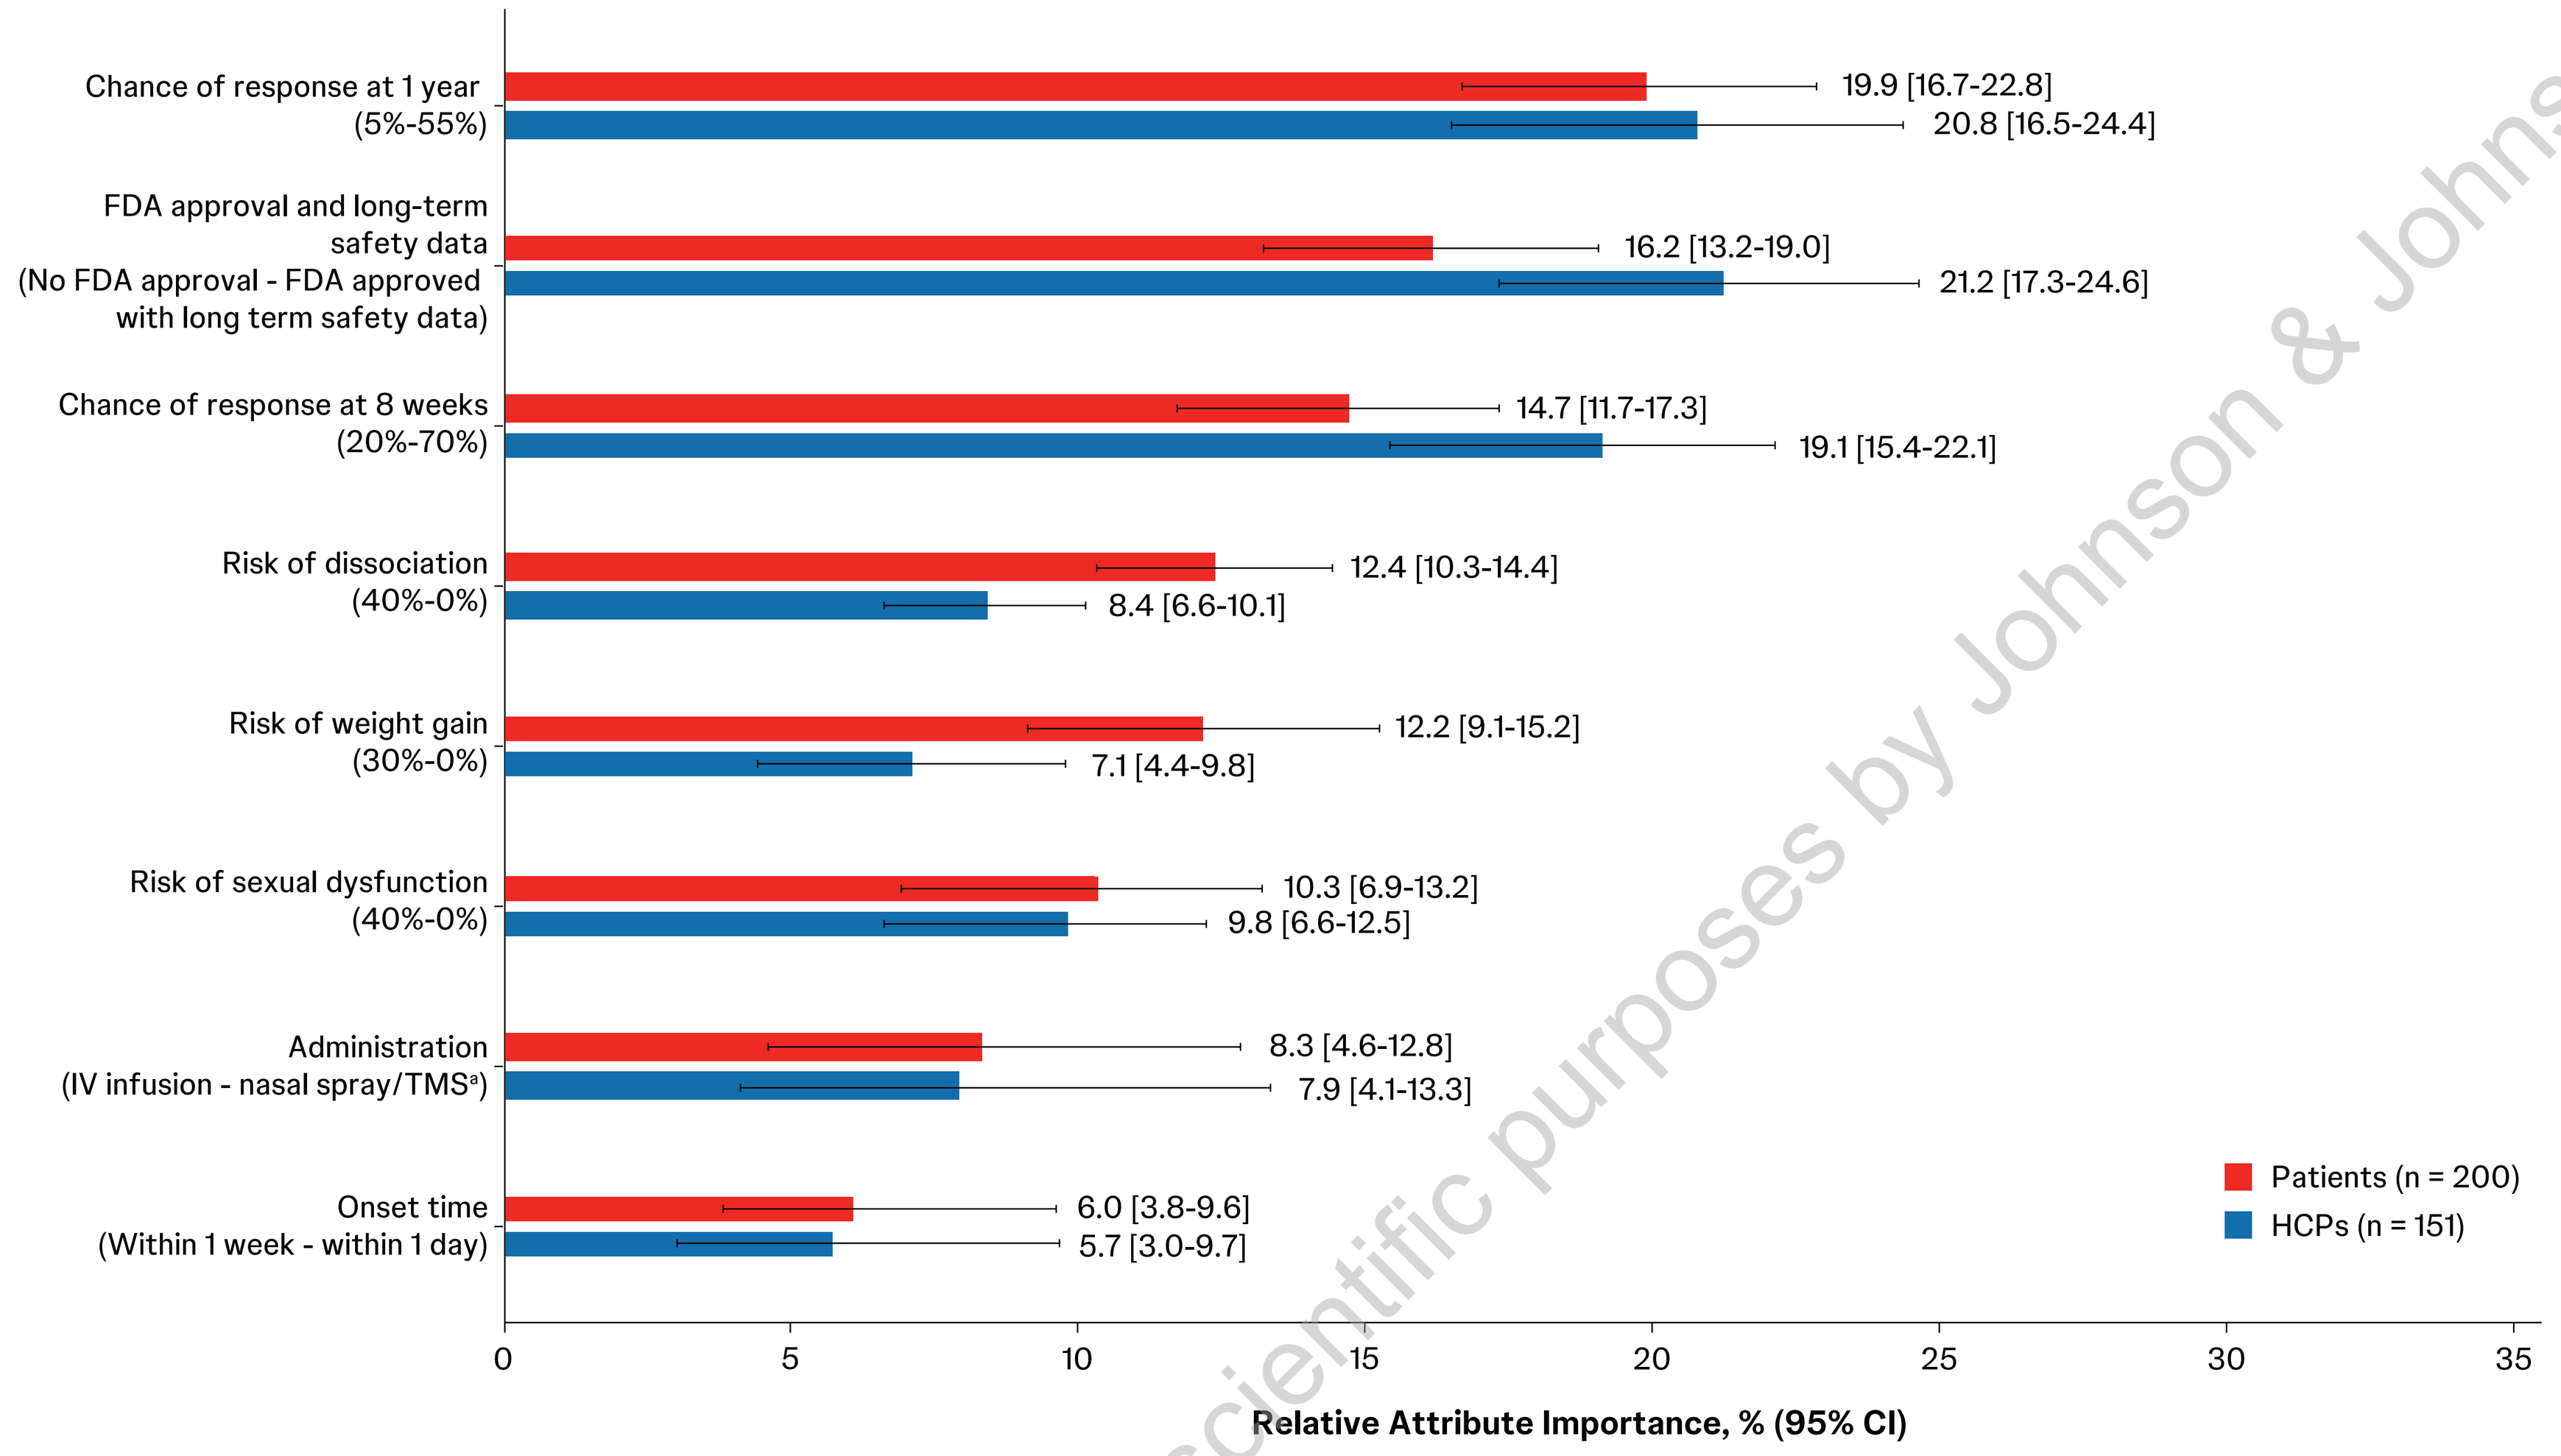
Chance of response at 1 year (5%–55%)
19.9 [16.7-22.8]
20.8 [16.5-24.4]
FDA approval and long-term safety data (No FDA approval - FDA approved with long term safety data)
16.2 [13.2-19.0]
21.2 [17.3-24.6]
Chance of response at 8 weeks (20%-70%)
14.7 [11.7-17.3]
19.1 [15.4-22.1]
Risk of dissociation (40%–0%)
12.4 [10.3-14.4]
8.4 [6.6-10.1]
Risk of weight gain (30%–0%)
12.2 [9.1-15.2]
7.1 [4.4-9.8]
Risk of sexual dysfunction (40%–0%)
10.3 [6.9-13.2]
9.8 [6.6-12.5]
Administration (IV infusion - nasal spray/TMS[a])
8.3 [4.6-12.8]
7.9 [4.1-13.3]
Onset time (Within 1 week - within 1 day)
6.0 [3.8-9.6]
5.7 [3.0-9.7]
0
5
10
15
20
25
30
35
Relative Attribute Importance, % (95% CI)
Patients (n = 200)
HCPs (n = 151)

DCE, discrete choice experiment; FDA, Food and Drug Administration; HCP, healthcare professional; IV, intravenous; RAI, relative attribute importance; TMS, transcranial magnetic stimulation.
The RAI was calculated using the utility difference between the least preferred and most preferred levels of each attribute included in the DCE (shown in brackets on the y-axis), with the exception of the RAI for risk of dissociation. The range included in the DCE for risk of dissociation was 80% to 0%; however, the RAI was based on an improvement from 40% to 0%, as this was considered to be a more clinically relevant improvement in risk of dissociation for the majority of TRD patients based on available data differentiating treatments.[9] Error bars represent 95% CIs around the RAI estimates.
[a]For patients, RAI for administration was calculated based on the utility difference between IV infusion (least preferred) and nasal spray (most preferred). For HCPs, RAI for administration was calculated based on the utility difference between IV infusion (least preferred) and TMS (most preferred).

Trade-offs: minimum acceptable increase in chance of 8-week response

- Both patients and HCPs were willing to trade improvements in 8-week response for stronger regulatory and safety evidence (**Figure 3**). To accept a treatment lacking FDA approval and long-term safety evidence, patients and HCPs required an approximately 55% (patients, 54.6%; HCPs, 56.1%) increase in 8-week response compared with a treatment supported by both FDA approval and long-term safety evidence. FDA approval without long-term safety evidence was valued as equivalent to a 40.4% increase in chance of 8-week response by patients and a 36.3% increase by HCPs
- Administration preferences were generally aligned across groups. Both patients and HCPs valued nasal spray similarly to oral treatment and preferred it over intravenous administration, requiring an approximately 17% improvement in 8-week response to accept intravenous treatment. Patients also preferred nasal spray over TMS, requiring a 30.0% improvement to accept TMS, while HCPs valued the 2 modes similarly ($P = 0.009$)
- Patients were more sensitive than HCPs to treatment-related risks. For each 10% reduction in dissociation risk, patients were willing to forgo a 10.7% chance of an 8-week response, compared with 5.5% among HCPs ($P < 0.001$). Eliminating weight-gain risk from 30% to 0% was valued as equivalent to a 43.1% chance of an 8-week response for patients and 19.2% for HCPs ($P = 0.004$)

Figure 3: Minimum acceptable increase in chance of response at 8 weeks

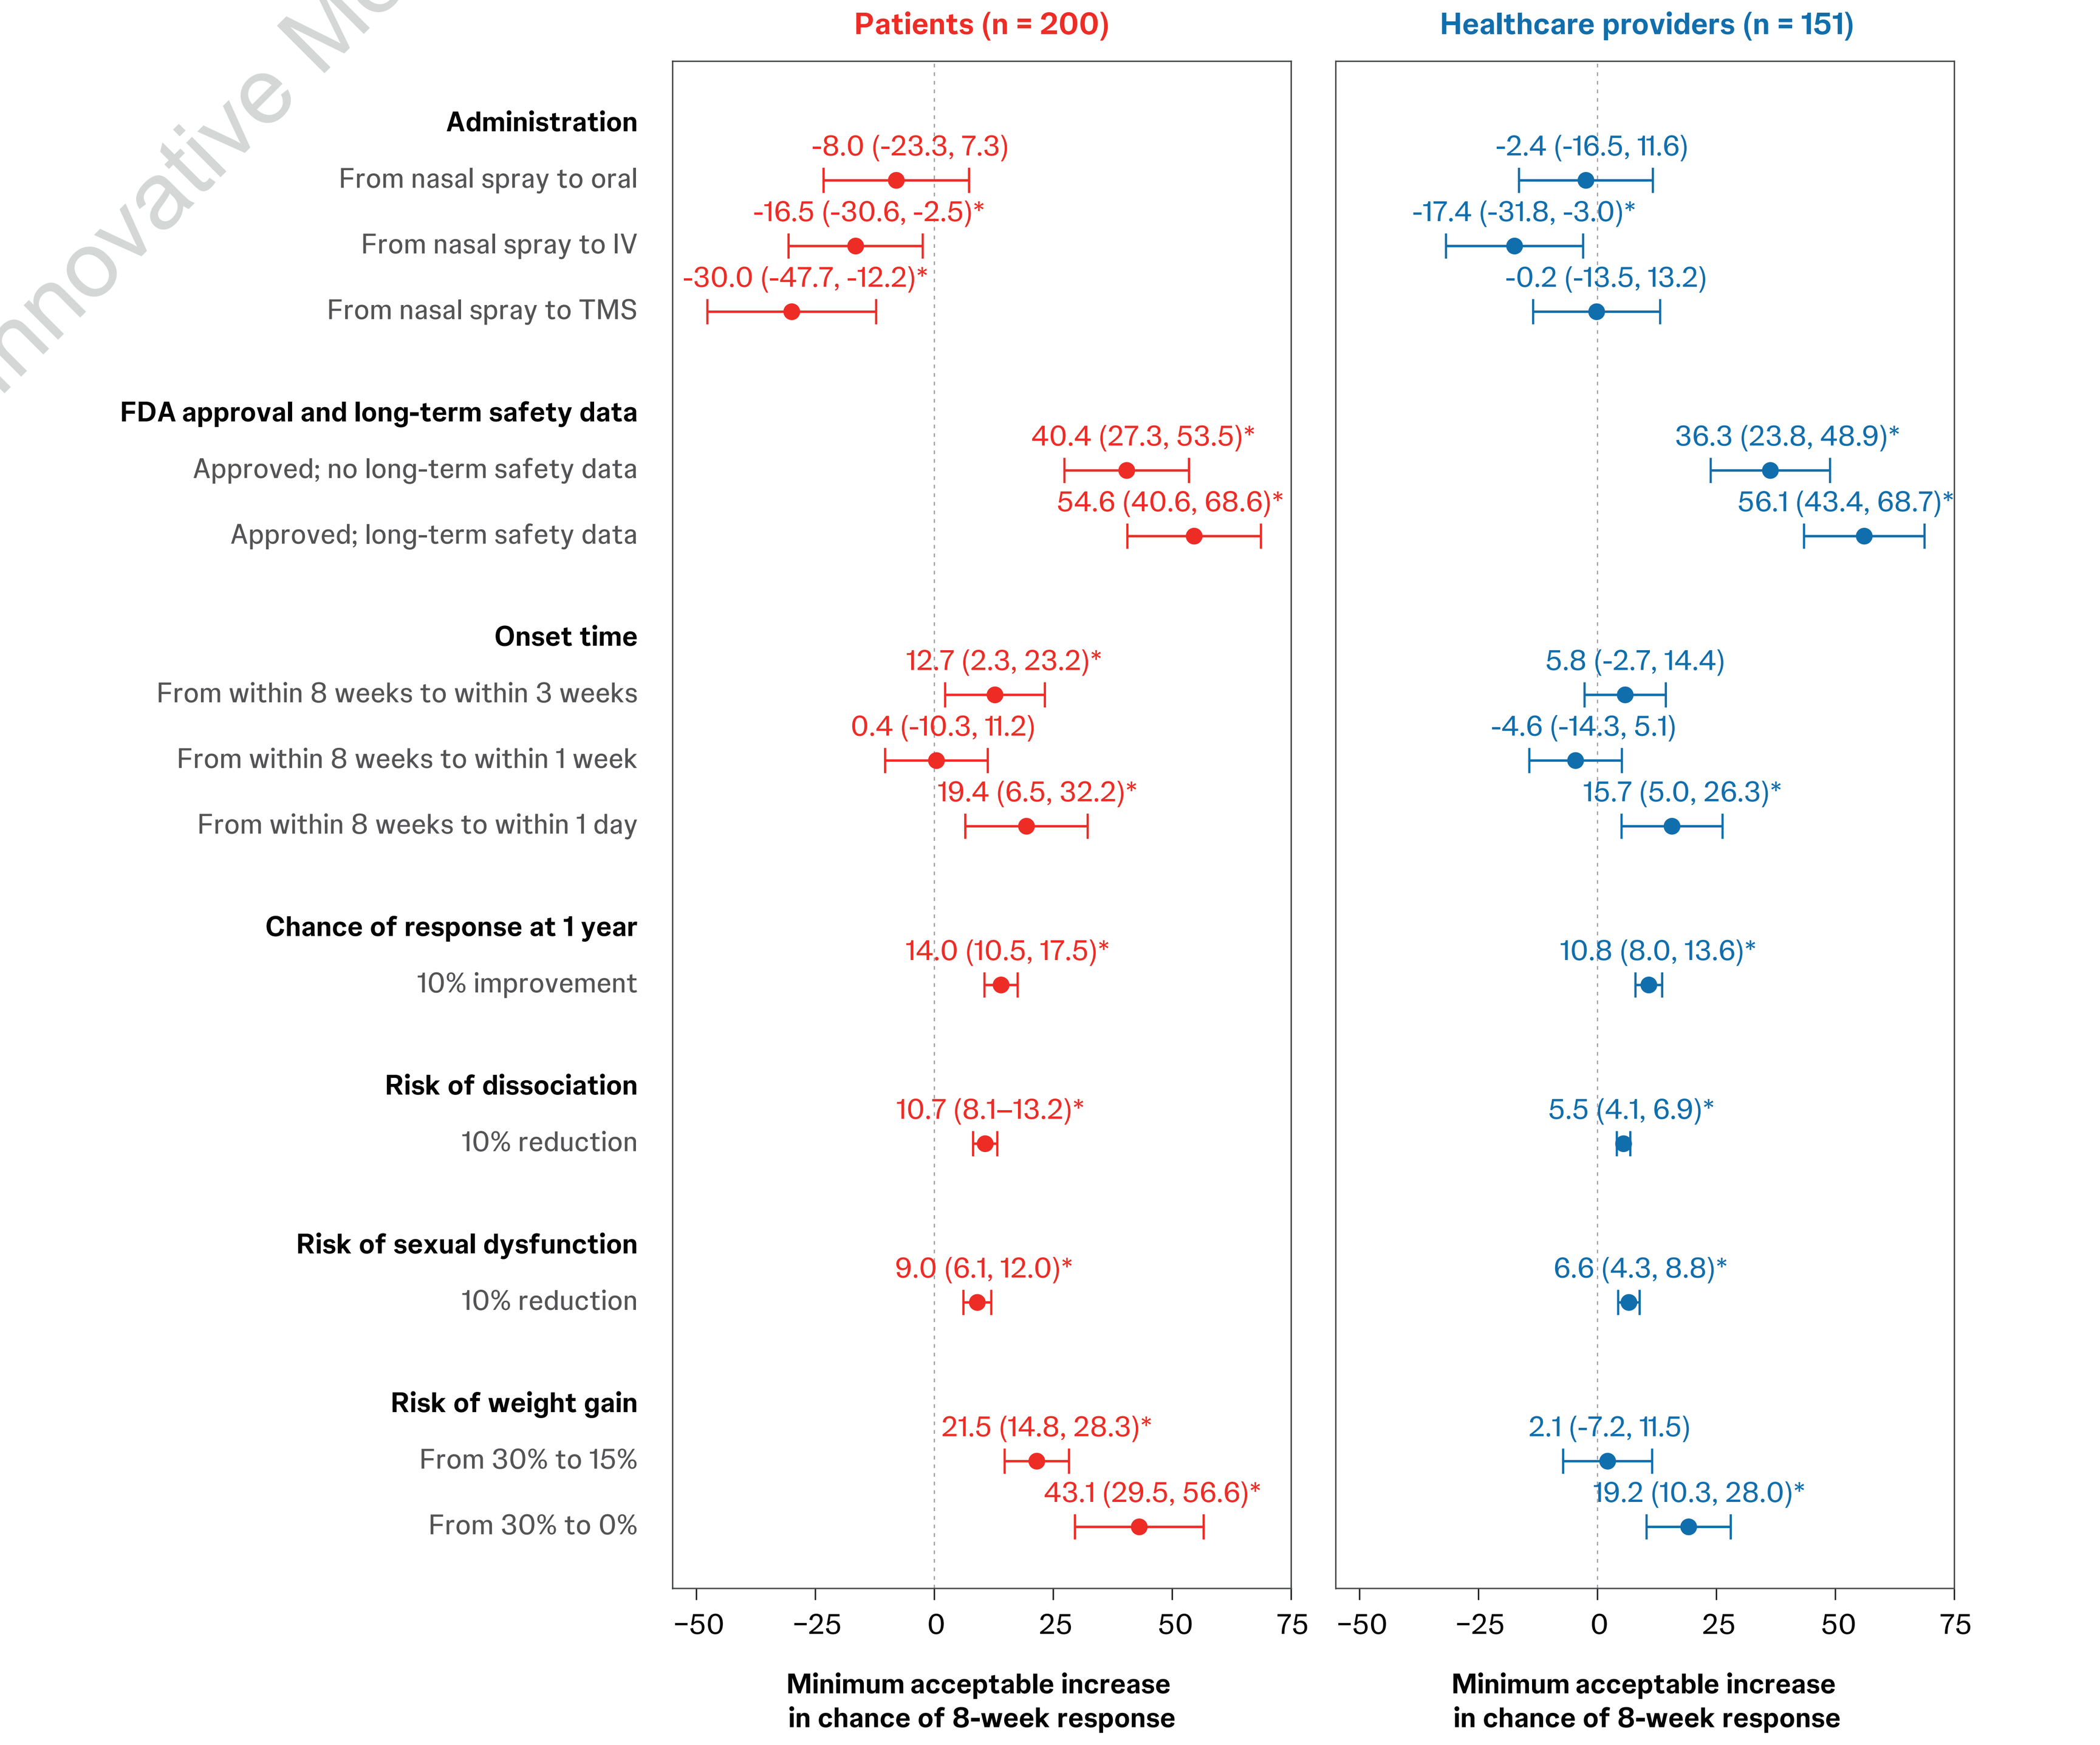
Patients (n = 200)
Healthcare providers (n = 151)
Administration
From nasal spray to oral
-8.0 (-23.3, 7.3)
-2.4 (-16.5, 11.6)
From nasal spray to IV
-16.5 (-30.6, -2.5)*
-17.4 (-31.8, -3.0)*
From nasal spray to TMS
-30.0 (-47.7, -12.2)*
-0.2 (-13.5, 13.2)
FDA approval and long-term safety data
Approved; no long-term safety data
40.4 (27.3, 53.5)*
36.3 (23.8, 48.9)*
Approved; long-term safety data
54.6 (40.6, 68.6)*
56.1 (43.4, 68.7)*
Onset time
From within 8 weeks to within 3 weeks
12.7 (2.3, 23.2)*
5.8 (-2.7, 14.4)
From within 8 weeks to within 1 week
0.4 (-10.3, 11.2)
-4.6 (-14.3, 5.1)
From within 8 weeks to within 1 day
19.4 (6.5, 32.2)*
15.7 (5.0, 26.3)*
Chance of response at 1 year
10% improvement
14.0 (10.5, 17.5)*
10.8 (8.0, 13.6)*
Risk of dissociation
10% reduction
10.7 (8.1–13.2)*
5.5 (4.1, 6.9)*
Risk of sexual dysfunction
10% reduction
9.0 (6.1, 12.0)*
6.6 (4.3, 8.8)*
Risk of weight gain
From 30% to 15%
21.5 (14.8, 28.3)*
2.1 (-7.2, 11.5)
From 30% to 0%
43.1 (29.5, 56.6)*
19.2 (10.3, 28.0)*
−50
−25
0
25
50
75
Minimum acceptable increase in chance of 8-week response

FDA, Food and Drug Administration; IV, intravenous; TMS, transcranial magnetic stimulation.
Significance: *P < 5%
Note on interpretation: Minimal acceptable increase in chance of 8-week response represents the additional chance of response at 8 weeks that participants would require to accept a less preferred treatment characteristic or, conversely, the amount of chance of 8-week response they would be willing to forgo to obtain a preferred treatment characteristic, relative to the specified baseline level. For example, patients required an additional 16.5% improvement in the chance of response at 8 weeks to choose IV infusion instead of intranasal treatment. Equivalently, patients were willing to forgo a 16.5% improvement in the chance of response at 8 weeks to receive intranasal treatment rather than IV infusion.

Preference heterogeneity by subgroups

- Patient preferences were generally consistent across subgroups, with only limited differences observed by time since diagnosis
- HCP preferences varied by profession, gender, and practice setting, particularly regarding response at 8 weeks, dissociation, and sexual dysfunction

Key Takeaways

- Patients and HCPs showed broadly aligned treatment preferences in TRD, with FDA approval and long-term safety evidence, efficacy, and treatment-related risks driving treatment choice
- Regulatory approval was a major driver of choice: Both patients and HCPs required an approximately 55% improvement in chance of 8-week response to accept treatments lacking FDA approval and long-term safety evidence, underscoring the importance of communicating the strength of the evidence base
- Administration influenced treatment choice: Although less influential than other preferences, patients and HCPs valued nasal spray similarly to oral treatment and preferred it over intravenous administration

Table 1: Study strengths and limitations

Strengths	Limitations
• Mixed-methods instrument development aligned with ISPOR good research practices • Confirmed MDD diagnosis and treatment history consistent with TRD for patient recruitment • Direct comparison of patients and HCPs using a common DCE • Strong implementation, validation, and choice-task engagement	• Stated hypothetical choices may differ from real-world decisions • US-only sample of predominantly women may limit generalizability • Patient sample had limited experience with ketamine, esketamine, or TMS • Subgroup analyses were exploratory, due to small subgroup sample sizes; no adjustment was made for multiple comparisons

DCE, discrete choice experiment; HCP, healthcare professional; ISPOR, The Professional Society for Health Economics and Outcomes Research; MDD, major depressive disorder; TMS, transcranial magnetic stimulation; TRD, treatment-resistant depression; US, United States.

Conclusions

- Patients and HCPs demonstrated broadly similar treatment preferences in TRD, with treatment choice driven primarily by response at 1 year, FDA approval with long-term safety evidence, and response at 8 weeks
- Greater patient sensitivity to dissociation and weight gain risks reinforces the importance of individualized benefit-risk discussions when selecting treatment for TRD

Acknowledgments

The authors thank Natasha Ramachandran for their support in conducting the cognitive pilot interviews and main survey research activities, and ApotheCom, Yardley, PA, for facilitating poster development.

Disclosures

JH, HL, LEP, and DA are employees of Thermo Fisher Scientific who received funding from Johnson & Johnson to support the study. JH is a Clinical Professor at Washington State University, a Senior Medical Science Liaison at Tempus ATI, and a key opinion leader for J&J. JAW is an employee and minority shareholder of Thermo Fisher Scientific who received funding from Johnson & Johnson to support the study. BB and DM are employees of Johnson & Johnson. RA has received honoraria for participation in the Johnson & Johnson Innovative Medicine speakers bureau for esketamine.

Novel Pathways in Depression

Scan the QR code

The QR code is intended to provide scientific information for individual reference, and the information should not be altered or reproduced in any way.

This study was funded by Johnson & Johnson.

Presented at Psych Congress; September 15-19, 2026; New Orleans, Louisiana, USA

References
1. Gaynes BN et al. *Depress Anxiety*. 2020;37(2):134-145. 2. Voineskos D et al. *Neuropsychiatr Dis Treat*. 2020;16:221-234. 3. Fairchild AO et al. *Neurol Psychiatry Brain Res.* 2020;37:67-78. 4. Fifer S et al. *Patient Prefer Adherence*. 2021;15:1621-1637. 5. Katz EG et al. *Ther Innov Regul Sci*. 2022;56(1):38-46. 6. Bridges JF et al. *Value Health*. 2011;14(4):403-413. 7. Clark MD et al. *Pharmacoeconomics*. 2014;32(9):883-902. 8. Soekhai V et al. *Pharmacoeconomics*. 2019;37(2):201-226. 9. SPRAVATO® (esketamine) nasal spray, CIII. Prescribing information. Janssen Pharmaceuticals, Inc.; 2025.

This material is distributed for scientific purposes by Johnson & Johnson Innovative Medicine and is not for promotional use.