94

Montgomery-Åsberg Depression Rating Scale Anhedonia Factor Score Following Esketamine Nasal Spray Monotherapy in Adult Patients With Treatment-Resistant Depression: A Post Hoc Analysis

Craig Chepke[1,2], Mai Himedan[3], Dong-Jing Fu[3], Oliver Lopena[3], Allen Wu[3], Ibrahim Turkoz[3], Lilianna Ly[3], Lisa Lim[3], Matthew Macaluso[4]

[1]Sandra & Leon Levine Psychiatry Residency Program at Atrium Health, Charlotte, NC; [2]Excel Psychiatric Associates, Huntersville, NC; [3]Johnson & Johnson, Titusville, NJ; [4]University of Alabama at Birmingham, Birmingham, AL

Introduction

- Anhedonia, defined as diminished interest or pleasure in response to stimuli that were previously perceived as rewarding, is experienced by approximately 70% of individuals diagnosed with major depressive disorder (MDD)[1]
- Anhedonia is a common residual symptom following treatment for MDD.[2] Residual symptoms that persist after treatment can predict higher depression relapse and recurrence rates and post-treatment functional impairment[3]
- Anhedonia is also a significant risk factor for suicide and suicidal ideation[4]
- In a real-world study in the United States (US), greater anhedonia severity was associated with reduced health-related quality of life, increased severity of depression and anxiety, greater impairment performing work and non–work-related activities, and higher direct medical costs[5]
- Esketamine nasal spray (ESK) in conjunction with an oral antidepressant was initially approved by the US Food and Drug Administration in 2019 for the treatment of treatment-resistant depression (TRD) in adults, and in 2020 for depressive symptoms in adults with MDD with acute suicidal ideation or behavior[6]
- A multicenter, double-blind, randomized, placebo-controlled, phase 4 study (NCT04599855) showed that ESK as monotherapy led to a rapid and superior improvement in Montgomery-Åsberg Depression Rating Scale (MADRS) total score compared with placebo at day 28[7]
 - Based on these results, ESK was approved by the US Food and Drug Administration in 2025 as the first and only monotherapy for adults with TRD[6]

Objective

- This post hoc analysis evaluated changes in anhedonia symptoms throughout 4 weeks of ESK monotherapy in patients with TRD

Methods

Study design

- This is a post hoc analysis of a multicenter, double-blind, randomized, placebo-controlled study (NCT04599855) conducted in the US (**Figure 1**)
- In the screening phase, patients were required to have taken no antidepressant medications for a minimum of 2 weeks directly before randomization
- In the double-blind treatment phase, patients were randomly assigned 1:1:2 to receive fixed doses of ESK 56 mg, ESK 84 mg, or placebo twice weekly for 4 weeks

Data analyses

- Efficacy was assessed in all randomly assigned patients who received ≥1 dose of study intervention in the double-blind treatment phase and who met predefined nonresponse criteria based on MADRS assessments during the screening phase (efficacy analysis set; **Figure 1**)
- The 5-item MADRS anhedonia factor score was used to measure anhedonia symptoms (range of score, 0-30) and is based on a subset of items (apparent sadness, reported sadness, concentration difficulties, lassitude, inability to feel) from the 10-item MADRS total score (range of item scores, 0-6)
- Change from baseline in the 5-item MADRS anhedonia factor score was assessed on days 2 (24 hours after treatment), 8, 15, 22, and 28 during the double-blind treatment phase
- The difference in mean change from baseline in depressive symptoms among patients treated with ESK (56 mg or 84 mg) versus placebo at day 28 was evaluated using analysis of covariance (ANCOVA) models, including treatment group, screening antidepressant status, and the baseline score as covariates
- A decrease from baseline, on average, in 5-item MADRS anhedonia factor score of ≥3 points was considered a clinically meaningful improvement, and a decrease of ≥6 points was considered a clinically substantial improvement[8,9]
 - These thresholds are derived by dividing the original published criteria based on the total 10-item MADRS total score by 2, as the anhedonia factor score encompasses 5 of the MADRS items
- The proportion of patients with a ≥3 point and ≥6 point improvement and a ≥50% improvement from baseline in 5-item MADRS anhedonia factor score were assessed. Treatment differences were evaluated using a Cochran–Mantel–Haenszel test adjusting for screening antidepressant status
- Safety was assessed in all randomly assigned patients who received ≥1 dose of study intervention in the double-blind treatment phase (safety analysis set)
- Treatment-emergent adverse events are summarized descriptively

Figure 1: Study design

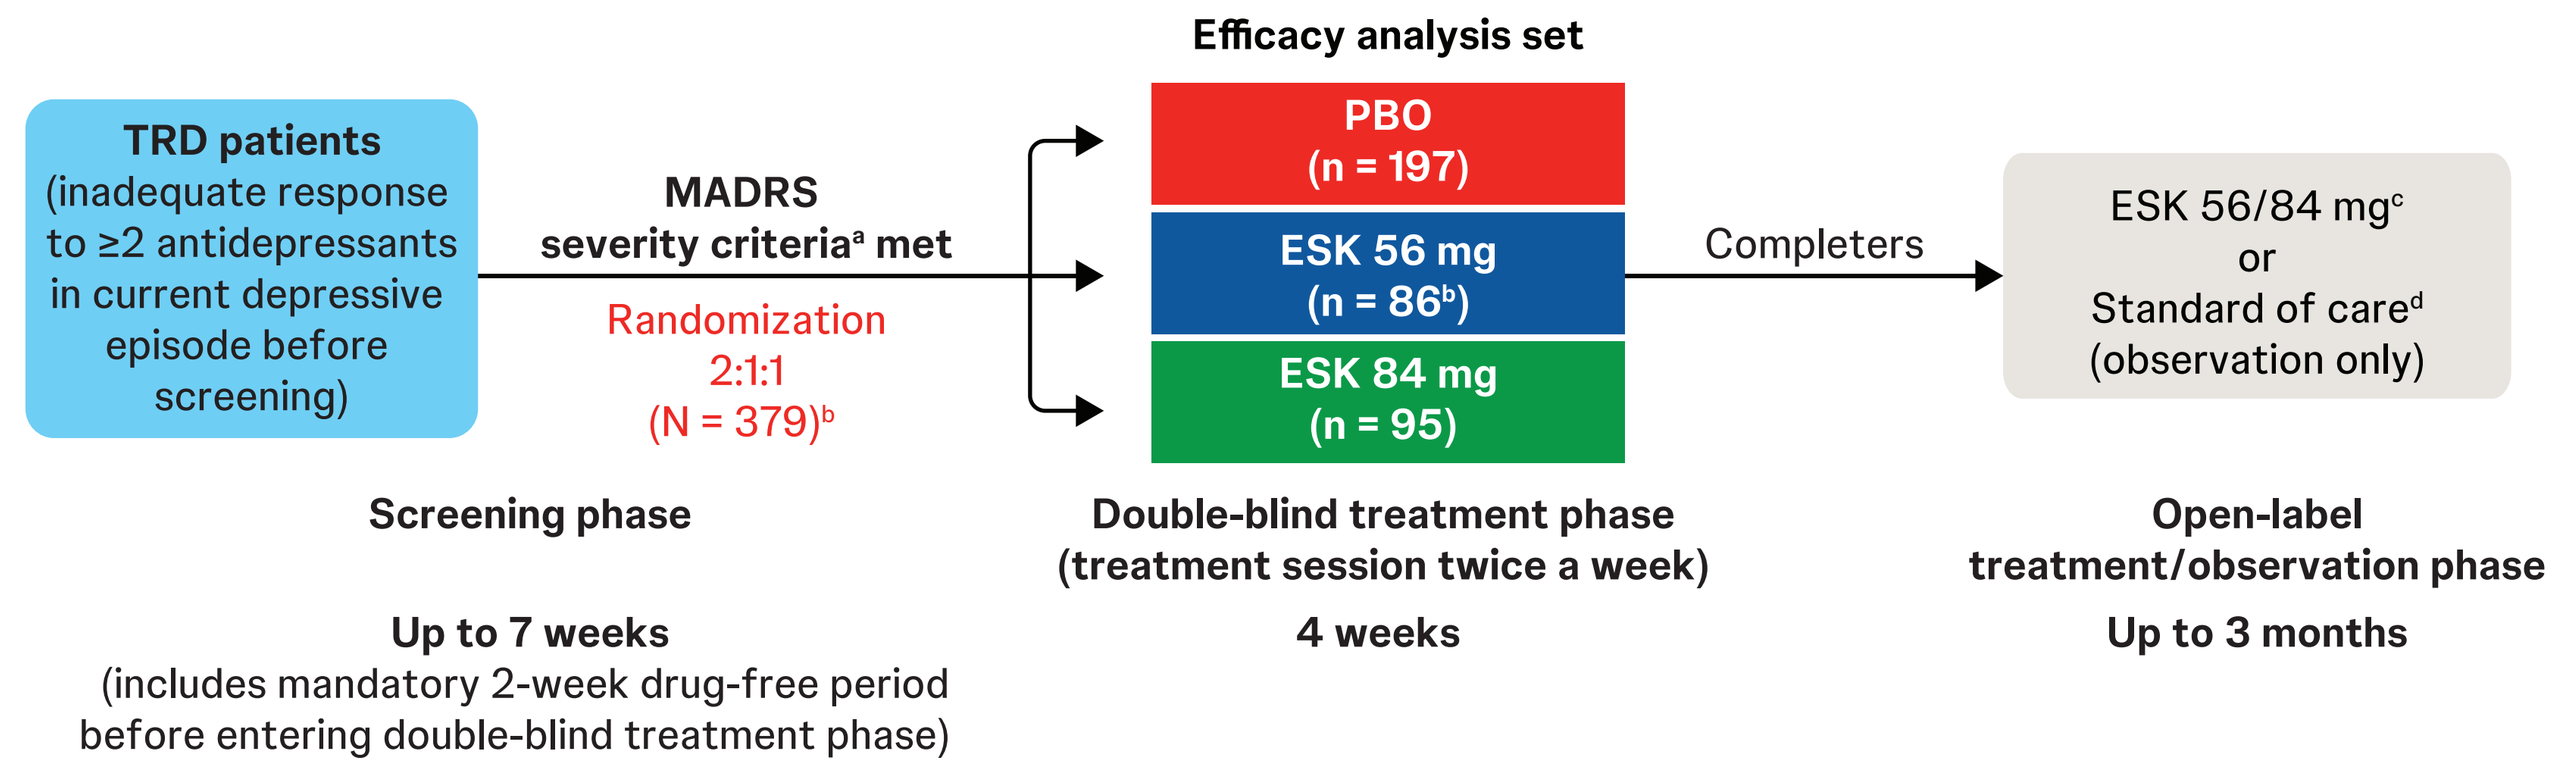
Efficacy analysis set
TRD patients
(inadequate response to ≥2 antidepressants in current depressive episode before screening)
MADRS severity criteria[a] met
Randomization
2:1:1
(N = 379)[b]
PBO
(n = 197)
ESK 56 mg
(n = 86[b])
ESK 84 mg
(n = 95)
Completers
ESK 56/84 mg[c]
or
Standard of care[d]
(observation only)
Screening phase
Up to 7 weeks
(includes mandatory 2-week drug-free period before entering double-blind treatment phase)
Double-blind treatment phase
(treatment session twice a week)
4 weeks
Open-label
treatment/observation phase
Up to 3 months

ESK, esketamine nasal spray; MADRS, Montgomery-Åsberg Depression Rating Scale; PBO, placebo; TRD, treatment-resistant depression.
Current antidepressant medications (including adjunctive treatments) were tapered during the screening phase, resulting in a ≥2-week antidepressant (and antipsychotic)-free observation period immediately before randomization.
[a]MADRS total score ≥28 at screening week 1, week 2, and day 1 (before randomization) and ≤25% improvement in the MADRS total score from screening week 1 to day 1 (before randomization); referred to as "nonresponse criteria" in the protocol.
[b]One of 87 patients randomly assigned to ESK 56 mg did not receive a dose of study drug and was not included in the efficacy analysis set.
[c]With or without standard of care.
[d]Only 1 patient received standard of care without ESK.

Results

Demographics and baseline characteristics

- A total of 378 adults with TRD met the predefined severity criteria and were randomly assigned to receive ESK 56 mg, ESK 84 mg, or placebo in the double-blind treatment phase (**Table 1**)

Table 1: Patient demographics and baseline clinical characteristics

	ESK 56 mg n = 86	ESK 84 mg n = 95	PBO n = 197	Total N = 378
Mean age (SD), years	46.5 (14.18)	44.8 (14.65)	45.2 (13.77)	45.4 (14.06)
Female, n (%)	51 (59.3)	61 (64.2)	119 (60.4)	231 (61.1)
Race, n (%)				
White	76 (88.4)	81 (85.3)	171 (86.8)	328 (86.8)
Black or African American	4 (4.7)	8 (8.4)	13 (6.6)	25 (6.6)
Other[a]	5 (5.8)	6 (6.3)	9 (4.6)	20 (5.3)
Mean duration of current episode (range), weeks	419.8 (12-2555)	406.4 (10-2236)	289.0 (10-1872)	348.3 (10-2555)
Number of episodes since diagnosis, n (%)				
≥3	54 (62.8)	55 (57.9)	127 (64.5)	236 (62.4)
Number of prior unsuccessful antidepressant trials, n (%)				
2	49 (57.0)	58 (61.1)	117 (59.4)	224 (59.3)
≥3	37 (43.0)	37 (38.9)	80 (40.6)	154 (40.7)
History of suicidal ideation in past 6 months, n (%)	38 (44.2)	52 (54.7)	105 (53.3)	195 (51.6)
Mean baseline CGI-S score (SD)	5.0 (0.60)	4.9 (0.65)	4.9 (0.61)	4.9 (0.62)
Mean baseline 10-item MADRS total score (SD)	37.5 (5.23)	36.6 (4.48)	37.5 (4.90)	37.3 (4.88)

CGI-S, Clinical Global Impression-Severity; ESK, esketamine nasal spray; MADRS, Montgomery-Åsberg Depression Rating Scale; PBO, placebo.
[a]Includes American Indian or Alaska Native, Asian, Native Hawaiian or Other Pacific Islander, Multiple. Does not include patients who were Not Reported (n = 4) or Unknown (n = 1).

- Most patients were female (61.1%) and White (86.8%), and patients had a mean (SD) age of 45.4 (14.06) years
- Patients had a mean (SD) Clinical Global Impression-Severity (CGI-S) score of 4.9 (0.62) and 10-item MADRS total score of 37.3 (4.88)

Change from baseline in 5-item MADRS anhedonia factor score

- At baseline, all treatment groups had a similar 5-item MADRS anhedonia factor score (**Table 2**)

Table 2: Baseline 5-item MADRS anhedonia factor score and individual item scores

	ESK 56 mg n = 86	ESK 84 mg n = 95	PBO n = 197
Mean 5-item MADRS anhedonia factor score (SD)	21.5 (2.78)	21.5 (2.56)	21.5 (2.86)
Mean MADRS anhedonia item scores (SD)			
Reported sadness	4.8 (0.66)	4.7 (0.71)	4.6 (0.72)
Apparent sadness	4.4 (0.83)	4.3 (0.80)	4.3 (1.00)
Concentration difficulties	4.1 (1.24)	3.9 (1.02)	4.0 (1.27)
Lassitude	4.2 (0.97)	4.3 (0.78)	4.2 (1.00)
Inability to feel	4.1 (0.82)	4.2 (0.83)	4.3 (0.72)

ESK, esketamine nasal spray; MADRS, Montgomery-Åsberg Depression Rating Scale; PBO, placebo.

- The LS mean change from baseline in 5-item MADRS anhedonia factor score was significantly greater in ESK 56 mg and ESK 84 mg groups compared with placebo at day 28 ($P < 0.001$) and as early as day 2 ($P < 0.01$; 24 hours post treatment; **Figure 2A**)
- At day 28, the LS mean difference (95% CI) from placebo in 5-item MADRS anhedonia factor scores for ESK 56 mg and ESK 84 mg were −3.4 (−5.38, −1.44) and −4.8 (−6.72, −2.89), respectively
- At day 28, all individual MADRS items scores (reported sadness, apparent sadness, concentration difficulties, lassitude, inability to feel) were significantly improved compared with placebo ($P < 0.05$; **Figure 2B**)

Improvements in 5-item MADRS anhedonia factor score

- Compared with placebo, significantly more ESK-treated patients had clinically meaningful improvement (≥3-point reduction) in 5-item MADRS anhedonia factor score at day 28 ($P < 0.001$) and day 2 ($P < 0.01$; 24 hours after treatment) and clinically substantial improvement (≥6-point reduction) at day 28 ($P < 0.01$) and day 2 ($P < 0.01$; 24 hours after treatment; **Figure 3**)
- Significantly more ESK-treated patients achieved ≥50% improvement from baseline in 5-item MADRS anhedonia factor score at day 28 (ESK 56 mg, 31.7%; ESK 84 mg, 33.7%, placebo, 16.2%; $P < 0.01$), and as early as day 2 ($P < 0.05$; **Figure 4**)

Safety

- In patients treated with ESK, the most commonly reported treatment-emergent adverse events were nausea (24.8%), dissociation (24.3%), dizziness (21.7%), and headache (19.0%), consistent with the established safety profile of ESK
- Most treatment-emergent adverse events occurred on dosing days, were mild or moderate in severity, and resolved on the same day
- Serious adverse events were reported in patients treated with ESK 56 mg (ankle fracture, n = 1), ESK 84 mg (suicide attempt, n = 1; ophthalmic migraine, n = 1), and placebo (self-injurious ideation, n = 1; suicidal ideation, n = 1; and myocardial infarction, n = 1), and none were considered related to study medication
- No deaths were reported

Figure 2: LS mean change from baseline in (A) 5-item MADRS anhedonia factor score over time and (B) individual MADRS item scores at day 28

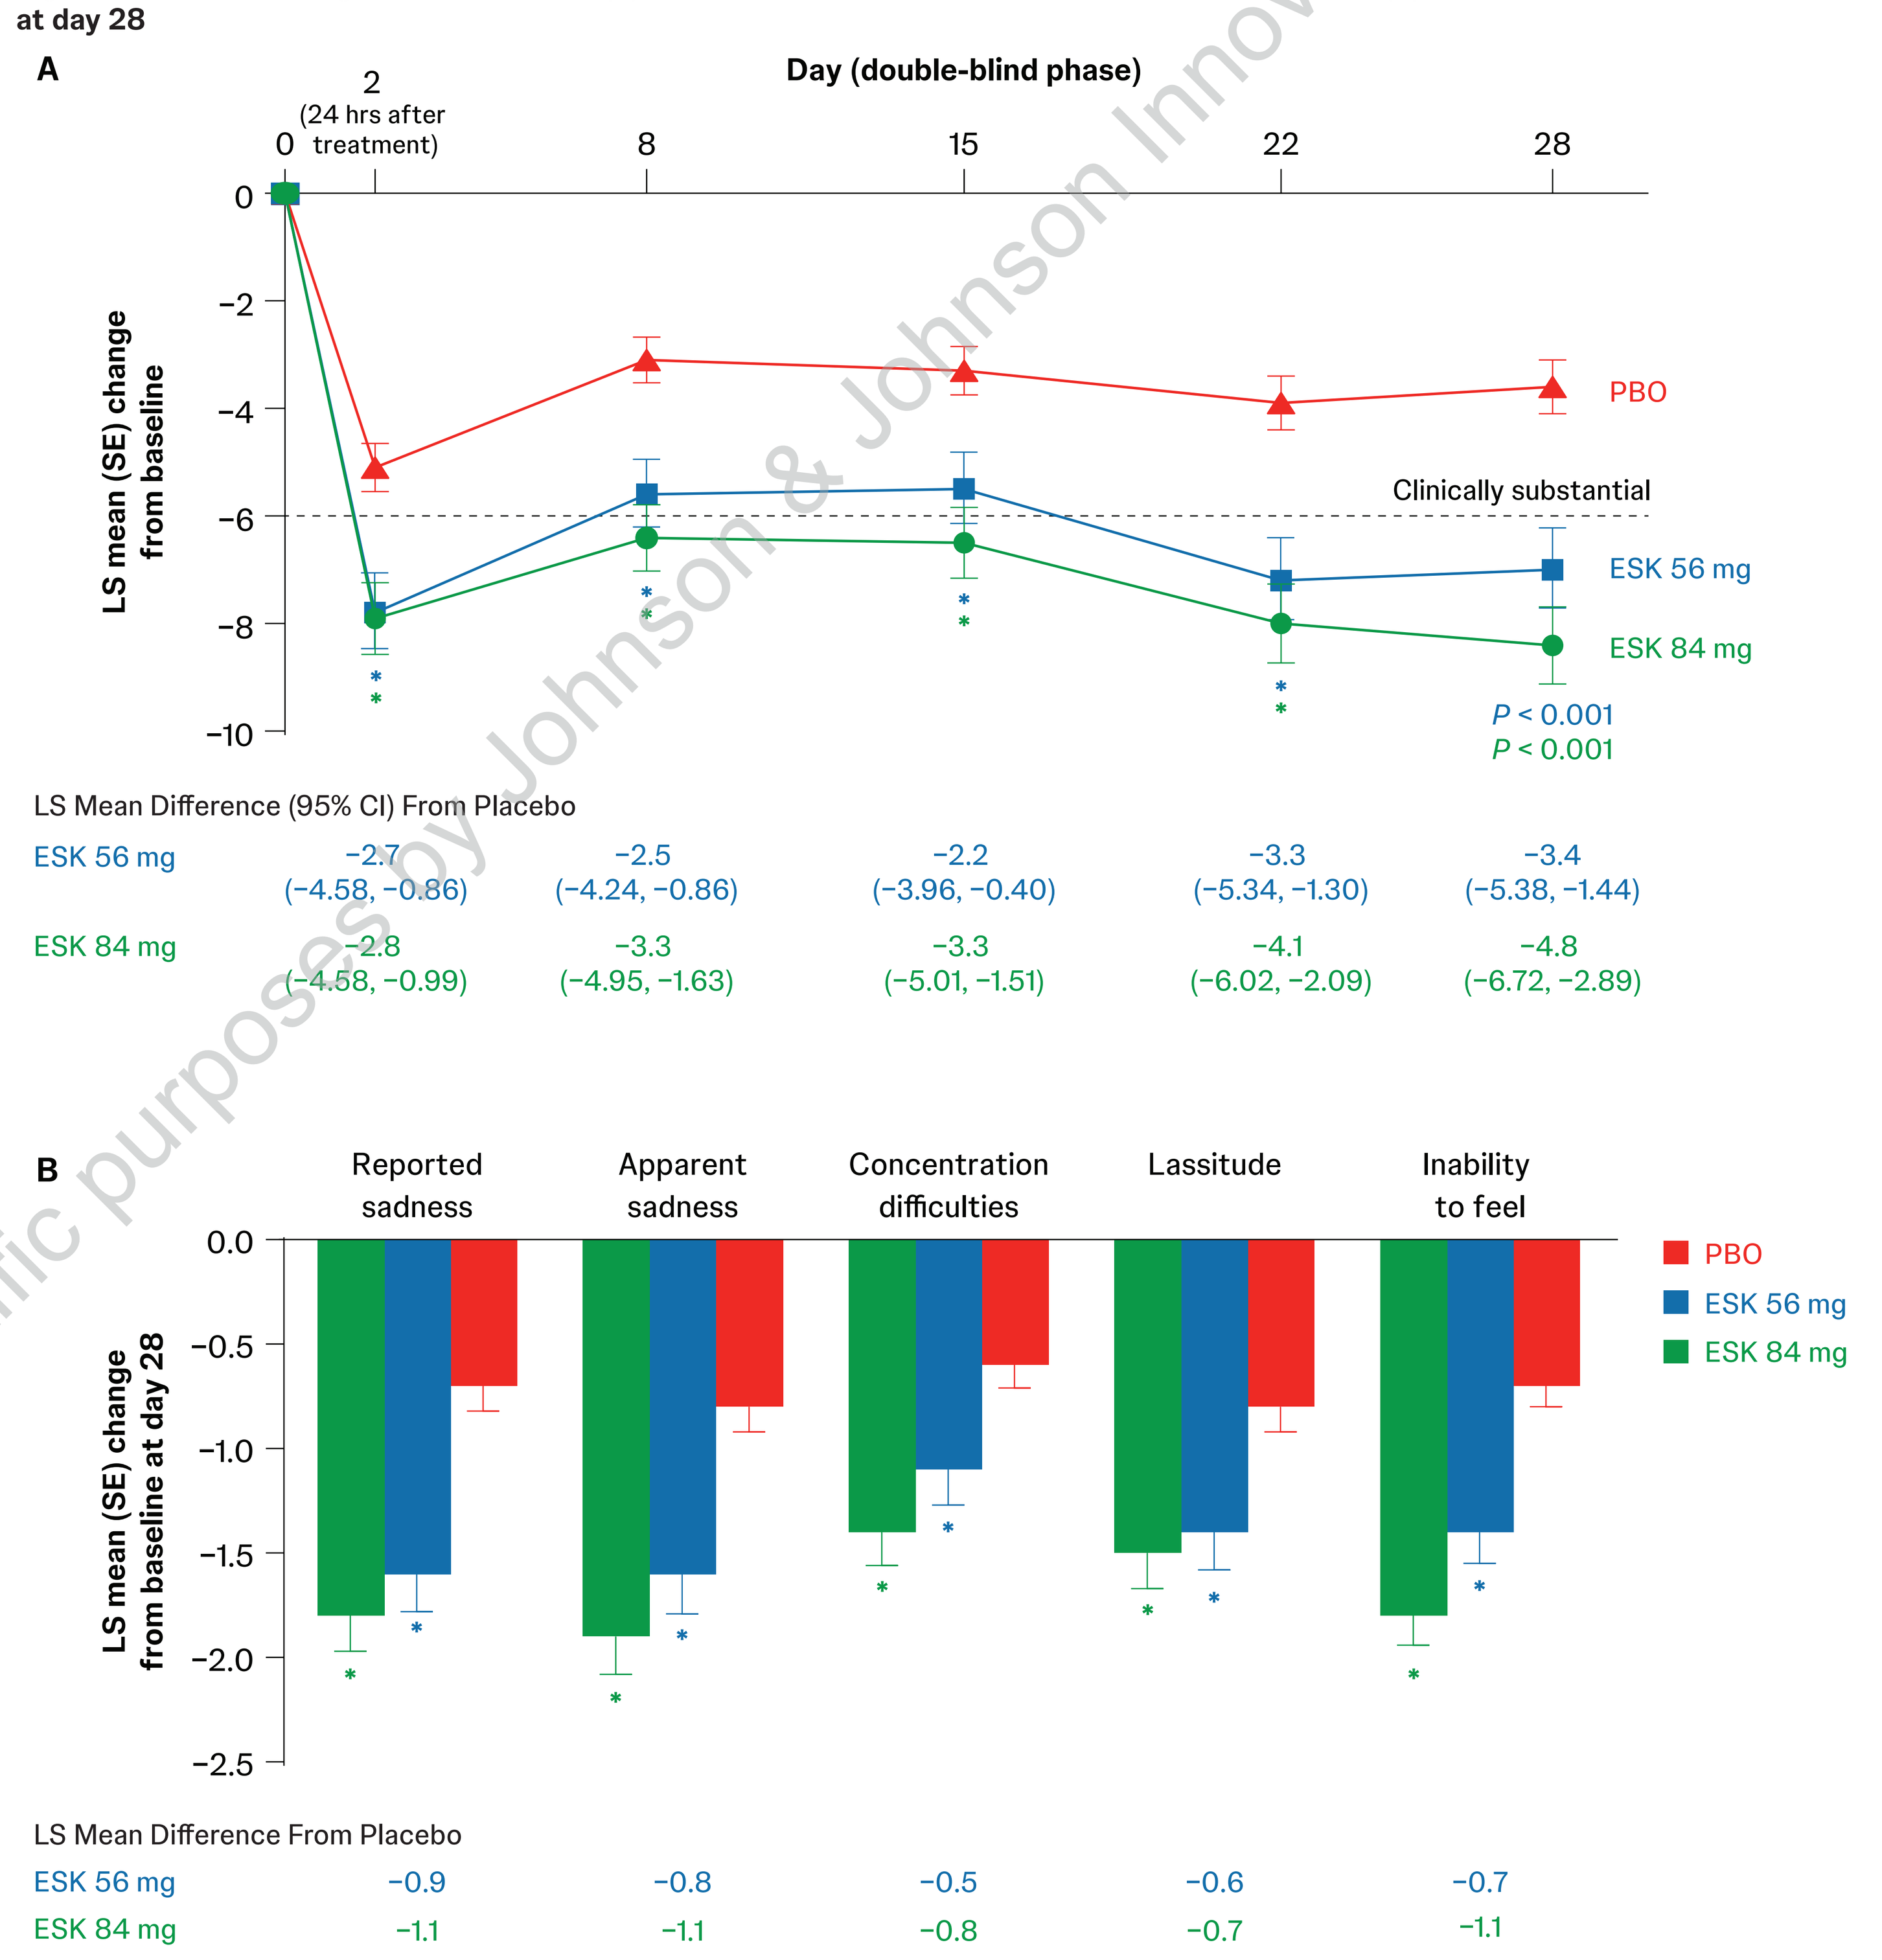
A
Day (double-blind phase)
0
2
(24 hrs after treatment)
8
15
22
28
LS mean (SE) change from baseline
0
−2
−4
−6
−8
−10
PBO
Clinically substantial
ESK 56 mg
ESK 84 mg
P < 0.001
P < 0.001
LS Mean Difference (95% CI) From Placebo
ESK 56 mg −2.7 (−4.58, −0.86) −2.5 (−4.24, −0.86) −2.2 (−3.96, −0.40) −3.3 (−5.34, −1.30) −3.4 (−5.38, −1.44)
ESK 84 mg −2.8 (−4.58, −0.99) −3.3 (−4.95, −1.63) −3.3 (−5.01, −1.51) −4.1 (−6.02, −2.09) −4.8 (−6.72, −2.89)
B
Reported sadness
Apparent sadness
Concentration difficulties
Lassitude
Inability to feel
LS mean (SE) change from baseline at day 28
0.0
−0.5
−1.0
−1.5
−2.0
−2.5
PBO
ESK 56 mg
ESK 84 mg
LS Mean Difference From Placebo
ESK 56 mg −0.9 −0.8 −0.5 −0.6 −0.7
ESK 84 mg −1.1 −1.1 −0.8 −0.7 −1.1

DB, double-blind; ESK, esketamine nasal spray; LS, least squares; MADRS, Montgomery-Åsberg Depression Rating Scale; PBO, placebo.
*$P < 0.05$, from analysis of covariance model compared with placebo.
Negative change in score indicates improvement in symptoms.

Figure 3: Proportion of patients with (A) ≥3-point or (B) ≥6-point improvement in 5-item MADRS anhedonia factor score

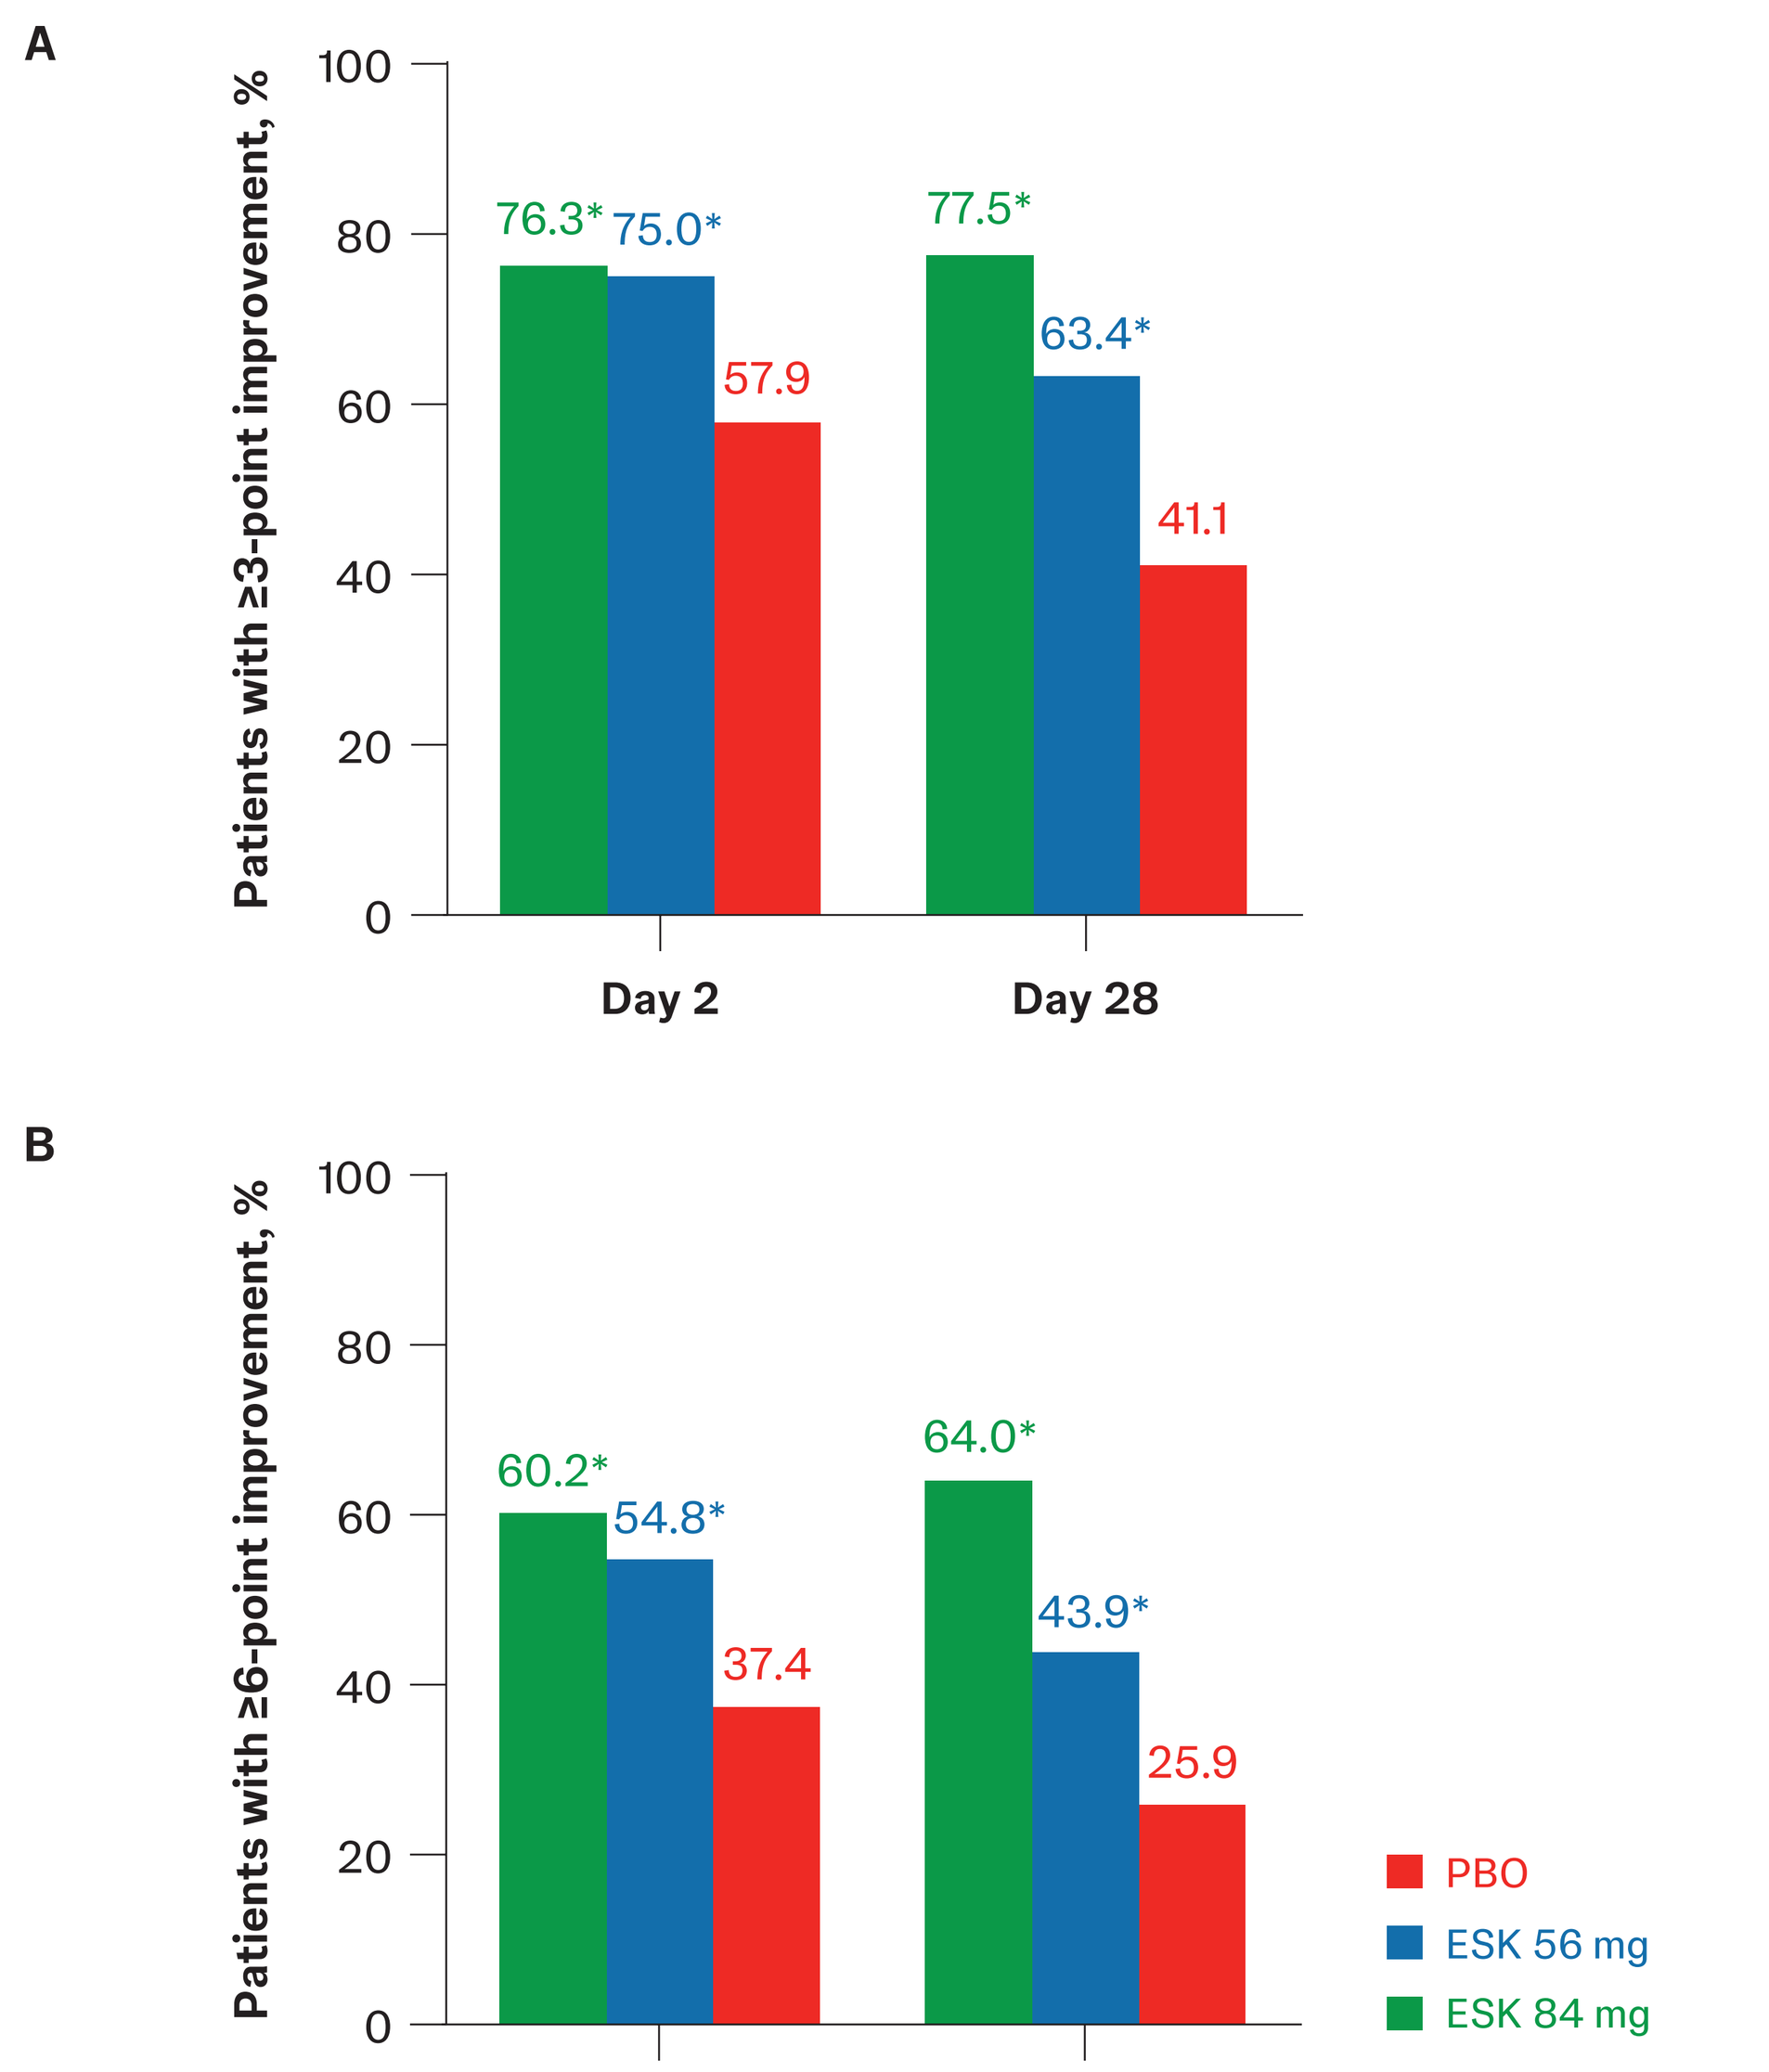
A
Patients with ≥3-point improvement, %
100
80
60
40
20
0
76.3*
75.0*
57.9
77.5*
63.4*
41.1
Day 2
Day 28
B
Patients with ≥6-point improvement, %
100
80
60
40
20
0
60.2*
54.8*
37.4
64.0*
43.9*
25.9
Day 2
Day 28
PBO
ESK 56 mg
ESK 84 mg

ESK, esketamine nasal spray; MADRS, Montgomery-Åsberg Depression Rating Scale; PBO, placebo.
*$P < 0.05$, from Cochran-Mantel-Haenszel test compared with placebo, adjusting for antidepressant status at study start.

Figure 4: Proportion of patients with ≥50% improvement from baseline in 5-item MADRS anhedonia factor score

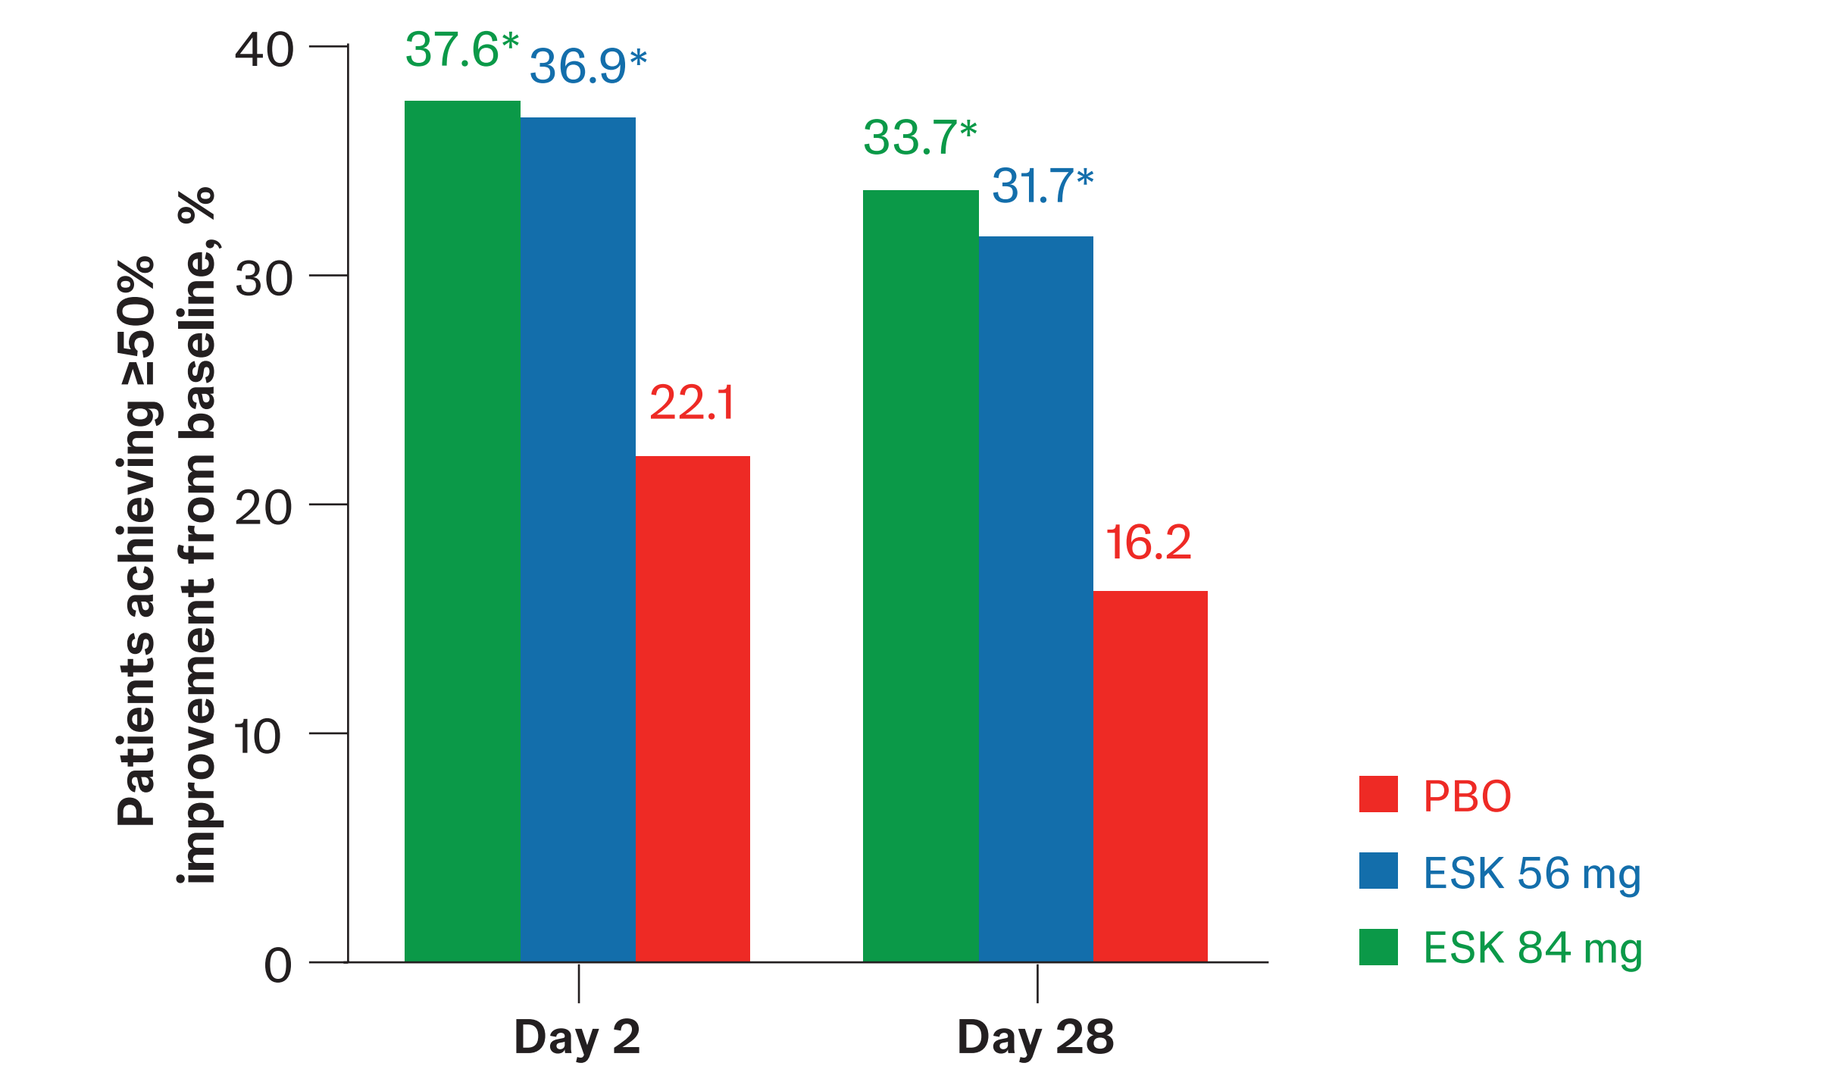
Patients achieving ≥50% improvement from baseline, %
40
30
20
10
0
37.6*
36.9*
22.1
33.7*
31.7*
16.2
Day 2
Day 28
PBO
ESK 56 mg
ESK 84 mg

ESK, esketamine nasal spray; MADRS, Montgomery-Åsberg Depression Rating Scale; PBO, placebo.
*$P < 0.05$, from Cochran-Mantel-Haenszel test compared with placebo, adjusting for antidepressant status at study start.

Key Takeaway

Esketamine nasal spray monotherapy led to rapid and robust improvements in anhedonia symptoms over 4 weeks of treatment, starting as early as 24 hours after first dose

Conclusions

In this post hoc analysis of a randomized, placebo-controlled trial, esketamine nasal spray monotherapy significantly improved 5-item MADRS anhedonia factor score and corresponding MADRS item scores at day 28 and as early as 24 hours after treatment compared with placebo in adults with treatment-resistant depression

Treatment-emergent adverse events were consistent with the established safety profile of ESK

Limitations

The 10-item MADRS is a widely used clinician-rated measure of depression severity, and while the 5-item MADRS anhedonia factor score and individual item scores have been previously used to measure changes in anhedonia symptoms after treatment, they are not specifically designed to measure anhedonia[8]

This is a post hoc analysis, which may limit the interpretation of the result

Acknowledgments

We dedicate this work to the memory of Dr. Vladimir Maletic, whose brilliance and generosity continue to inspire us.

The authors thank Emily Spaulding, PhD (ApotheCom, Yardley, PA), for editorial and writing assistance, which was funded by Johnson & Johnson.

Disclosures

CC has served on an advisory board for AbbVie, Acadia, Alkermes, Axsome, Biogen, Bristol Myers Squibb, Corium, Eli Lilly, Idorsia, Intra-Cellular, Jazz, Johnson & Johnson, Lundbeck, Moderna, Neurocrine, Otsuka, Sage, Sumitomo, Takeda, and Teva; has a spouse who served on an advisory board for Bristol Myers Squibb and Otsuka; has served in a consulting role for AbbVie, Acadia, Alkermes, Axsome, Biogen, Boehringer Ingelheim, Bristol Myers Squibb, Corium, Eli Lilly, Intra-Cellular, Johnson & Johnson, Lundbeck, MedinCell, Moderna, Neurocrine, Otsuka, Sage, Sumitomo, Supernus, and Teva; has received research funding/grant support from Acadia, Axsome, Harmony, Neurocrine, and Teva; and has served on a speaker's bureau for AbbVie, Acadia, Alkermes, Axsome, Boehringer Ingelheim, Bristol Myers Squibb, Corium, Intra-Cellular, Jazz, Johnson & Johnson, Lundbeck, Luye, Merck, Neurocrine, Otsuka, Sumitomo, Takeda, and Teva.
MH, D-JF, OL, AW, IB, L Ly, and L Lim are employees of Johnson & Johnson and may hold stock or restricted stock units in Johnson & Johnson.
MM has conducted clinical research as principal investigator over the last year for Acadia, Allergan, Alkermes, AssureRx/Myriad, Eisai, Lundbeck, Janssen, Neurim, SAGE Pharmaceuticals, and Suven. All clinical trial and study contracts were with and payments made to the Kansas University Medical Center Research Institute, a research institute affiliated with Kansas University School of Medicine-Wichita. MM is a member of the speaker bureau for Johnson & Johnson.

Previously presented at Psych Congress; September 17-21, 2025; San Diego, California, USA

Novel Pathways in Depression

Scan the QR code

The QR code is intended to provide scientific information for individual reference, and the information should not be altered or reproduced in any way.

This study was funded by Johnson & Johnson.

Presented at Psych Congress; September 15-19, 2026; New Orleans, Louisiana, USA

References

1. Cao B, et al. *Prog Neuropsychopharmacol Biol Psychiatry*. 2019;92:109-117. 2. Whiston A, et al. *Front Psychiatry*. 2022;13:746678. 3. Wojnarowski C, et al. *Behav Cogn Psychother*. 2019;47(5):514-529. 4. Whitton AE, Pizzagalli DA. *Curr Top Behav Neurosci*. 2022;58:111-127. 5. Kale H, et al. *Int J Neuropsychopharmacol*. 2025;28(8):pyaf048. 6. SPRAVATO® (esketamine) nasal spray, CIII. Prescribing information. Janssen Pharmaceuticals, Inc.; 2026:16. Accessed July 7, 2026. https://www.janssenlabels.com/package-insert/product-monograph/prescribing-information/SPRAVATO-pi.pdf 7. Janik A, et al. *JAMA Psychiatry*. 2025. Published online. 8. McIntyre RS, et al. *J Affect Disord*. 2024;363:430-435. 9. Turkoz I, et al. *Acta Psychiatr Scand*. 2021;143(3):253-263.

This material is distributed for scientific purposes by Johnson & Johnson Innovative Medicine and is not for promotional use.