83

Incremental Healthcare Resource Utilization and Costs Associated With Depression Severity Measured by the Patient Health Questionnaire-9

Dejan Milentijevic[1], Arthur Voegel[2], Maryia Zhdanava[3], Dominic Pilon[3], Anabelle Tardif-Samson[3], Yuxi Wang[3], Brahim Bookhart[1], Paul E Greenberg[4]

[1]Johnson & Johnson, Titusville, NJ, USA; [2]Analysis Group, Inc., New York, NY, USA; [3]Analysis Group, Inc., Montréal, QC, Canada; [4]Analysis Group, Inc., Boston, MA, USA

Introduction

- Depression is a common, disabling condition associated with substantial healthcare resource utilization (HRU), medical expenditures, and productivity loss[1]
- However, limited evidence has quantified how HRU and costs vary by depression severity
- This study linked patient-reported Patient Health Questionnaire-9 (PHQ-9) scores with claims data to quantify the incremental economic burden associated with increasing depression severity

Methods

Data source

- Closed claims data from Komodo Research Data (KRD+) were used (January 1, 2022, to December 31, 2025)
- Data were de-identified and complied with the patient requirements of the Health Insurance Portability and Accountability Act

Study design

- A retrospective observational design was used
- The index date was defined as 60 days prior to a randomly selected PHQ-9 score
- Baseline and follow-up included the 12 months of continuous insurance eligibility before and after the index date, respectively; follow-up was censored at the first change in PHQ-9 severity category

Study population

- Patients met the following selection criteria:
 - ≥1 PHQ-9 score
 - ≥12 months of continuous insurance eligibility before and after the index date
 - ≥18 years old on the index date

Measures and outcomes

- The PHQ-9 is a 9-item questionnaire assessing depression severity over the past 2 weeks; scores range from 0 to 27, with higher scores indicating greater severity[2]
 - The severity categories were none/minimal (0-4), mild (5-9), moderate (10-14), moderately severe (15-19), and severe (20-27)
- Annual all-cause HRU and costs (medical and pharmacy costs) were assessed; costs were adjusted for inflation to 2025 US dollars using the US Consumer Price Index

Statistical analysis

- Adjusted models estimated annual incremental HRU and costs during follow-up by PHQ-9 severity category versus none/minimal depression and per 5-point increase in PHQ-9 score (i.e., minimal clinically important difference)
 - HRU outcomes were evaluated using Poisson generalized linear models (GLMs) and reported as rate ratios
 - Costs were evaluated using ordinary least squares (OLS) regression and reported as adjusted mean differences
- Models were adjusted for baseline characteristics and comorbidities

Results

Patient characteristics

- The overall cohort included 978,540 patients; mean age 54.2 years; 59.8% female; baseline characteristics are reported in **Table 1**
 - Greater depression severity was associated with younger age, more obesity, pulmonary disease, and psychiatric diagnoses, while hypertension was more common in the none/minimal group

Table 1: Baseline patient characteristics overall and by PHQ-9 depression severity

Mean ± SD [median] or n (%)	Overall	None/minimal (0-4)	Mild (5-9)	Moderate (10-14)	Moderately severe (15-19)	Severe (20-27)
	N = 978,540	N = 740,129	N = 128,021	N = 61,376	N = 32,505	N = 16,509
Age at index date, years	54.2 ± 19.1 [56.0]	55.8 ± 18.8 [58.0]	51.0 ± 19.4 [51.0]	47.8 ± 19.3 [46.0]	46.0 ± 18.7 [44.0]	44.7 ± 17.6 [43.0]
Female	585,272 (59.8)	427,007 (57.7)	83,474 (65.2)	41,221 (67.2)	22,295 (68.6)	11,275 (68.3)
Race/Ethnicity						
White	686,558 (70.2)	522,857 (70.6)	90,638 (70.8)	41,527 (67.7)	21,147 (65.1)	10,389 (62.9)
Racial minority	272,456 (27.8)	202,968 (27.4)	34,601 (27.0)	18,472 (30.1)	10,656 (32.8)	5,759 (34.9)
Unknown	19,526 (2.0)	14,304 (1.9)	2,782 (2.2)	1,377 (2.2)	702 (2.2)	361 (2.2)
Payer type						
Commercial	453,836 (46.4)	346,321 (46.8)	59,848 (46.7)	27,963 (45.6)	13,581 (41.8)	6,123 (37.1)
Medicare Advantage	361,872 (37.0)	291,390 (39.4)	41,276 (32.2)	17,125 (27.9)	8,260 (25.4)	3,821 (23.1)
Medicaid	148,728 (15.2)	91,524 (12.4)	25,141 (19.6)	15,480 (25.2)	10,251 (31.5)	6,332 (38.4)
Unknown	14,104 (1.4)	10,894 (1.5)	1,756 (1.4)	808 (1.3)	413 (1.3)	233 (1.4)
CCI score	1.0 ± 1.6 [0.0]	1.0 ± 1.6 [0.0]	1.0 ± 1.7 [0.0]	1.0 ± 1.7 [0.0]	1.0 ± 1.7 [0.0]	0.9 ± 1.6 [0.0]
Selected physical comorbidities						
Hypertension	376,551 (38.5)	295,509 (39.9)	45,350 (35.4)	20,306 (33.1)	10,301 (31.7)	5,085 (30.8)
Obesity	195,666 (20.0)	140,147 (18.9)	29,350 (22.9)	14,520 (23.7)	7,732 (23.8)	3,917 (23.7)
Chronic pulmonary disease	140,257 (14.3)	98,537 (13.3)	21,270 (16.6)	11,140 (18.2)	6,065 (18.7)	3,245 (19.7)
Selected DSM-5 diagnoses						
Anxiety disorders	229,388 (23.4)	138,326 (18.7)	44,131 (34.5)	24,846 (40.5)	14,231 (43.8)	7,854 (47.6)
Depressive disorders	200,564 (20.5)	110,949 (15.0)	41,939 (32.8)	24,899 (40.6)	14,645 (45.1)	8,132 (49.3)
Sleep-wake disorders	175,014 (17.9)	118,484 (16.0)	29,442 (23.0)	15,047 (24.5)	8,001 (24.6)	4,040 (24.5)
Baseline antidepressant use	312,213 (31.9)	194,728 (26.3)	58,277 (45.5)	31,945 (52.0)	17,844 (54.9)	9,419 (57.1)

CCI, Charlson Comorbidity Index; DSM-5, Diagnostic and Statistical Manual of Mental Disorders, Fifth Edition; PHQ-9, Patient Health Questionnaire-9; SD, standard deviation.

Healthcare resource utilization

- Patients with none/minimal depression averaged 0.1 inpatient (IP) admissions, 1.4 IP days, 0.7 emergency department (ED) visits, and 19.6 outpatient (OP) visits annually
 - Relative to none/minimal depression, annual all-cause HRU gradually increased across mild to severe depression by 9% to 78% for IP admissions, 28% to 149% for IP days, 10% to 61% for ED visits, and 15% to 48% for OP visits, respectively (**Figure 1**)
- Each 5-point PHQ-9 increase was associated with 14% more IP admissions, 27% more IP days, 12% more ED visits, and 11% more OP visits (**Figure 2**)

Figure 1: Adjusted rate ratios of annual all-cause HRU by PHQ-9 depression severity relative to "none/minimal" category[a]

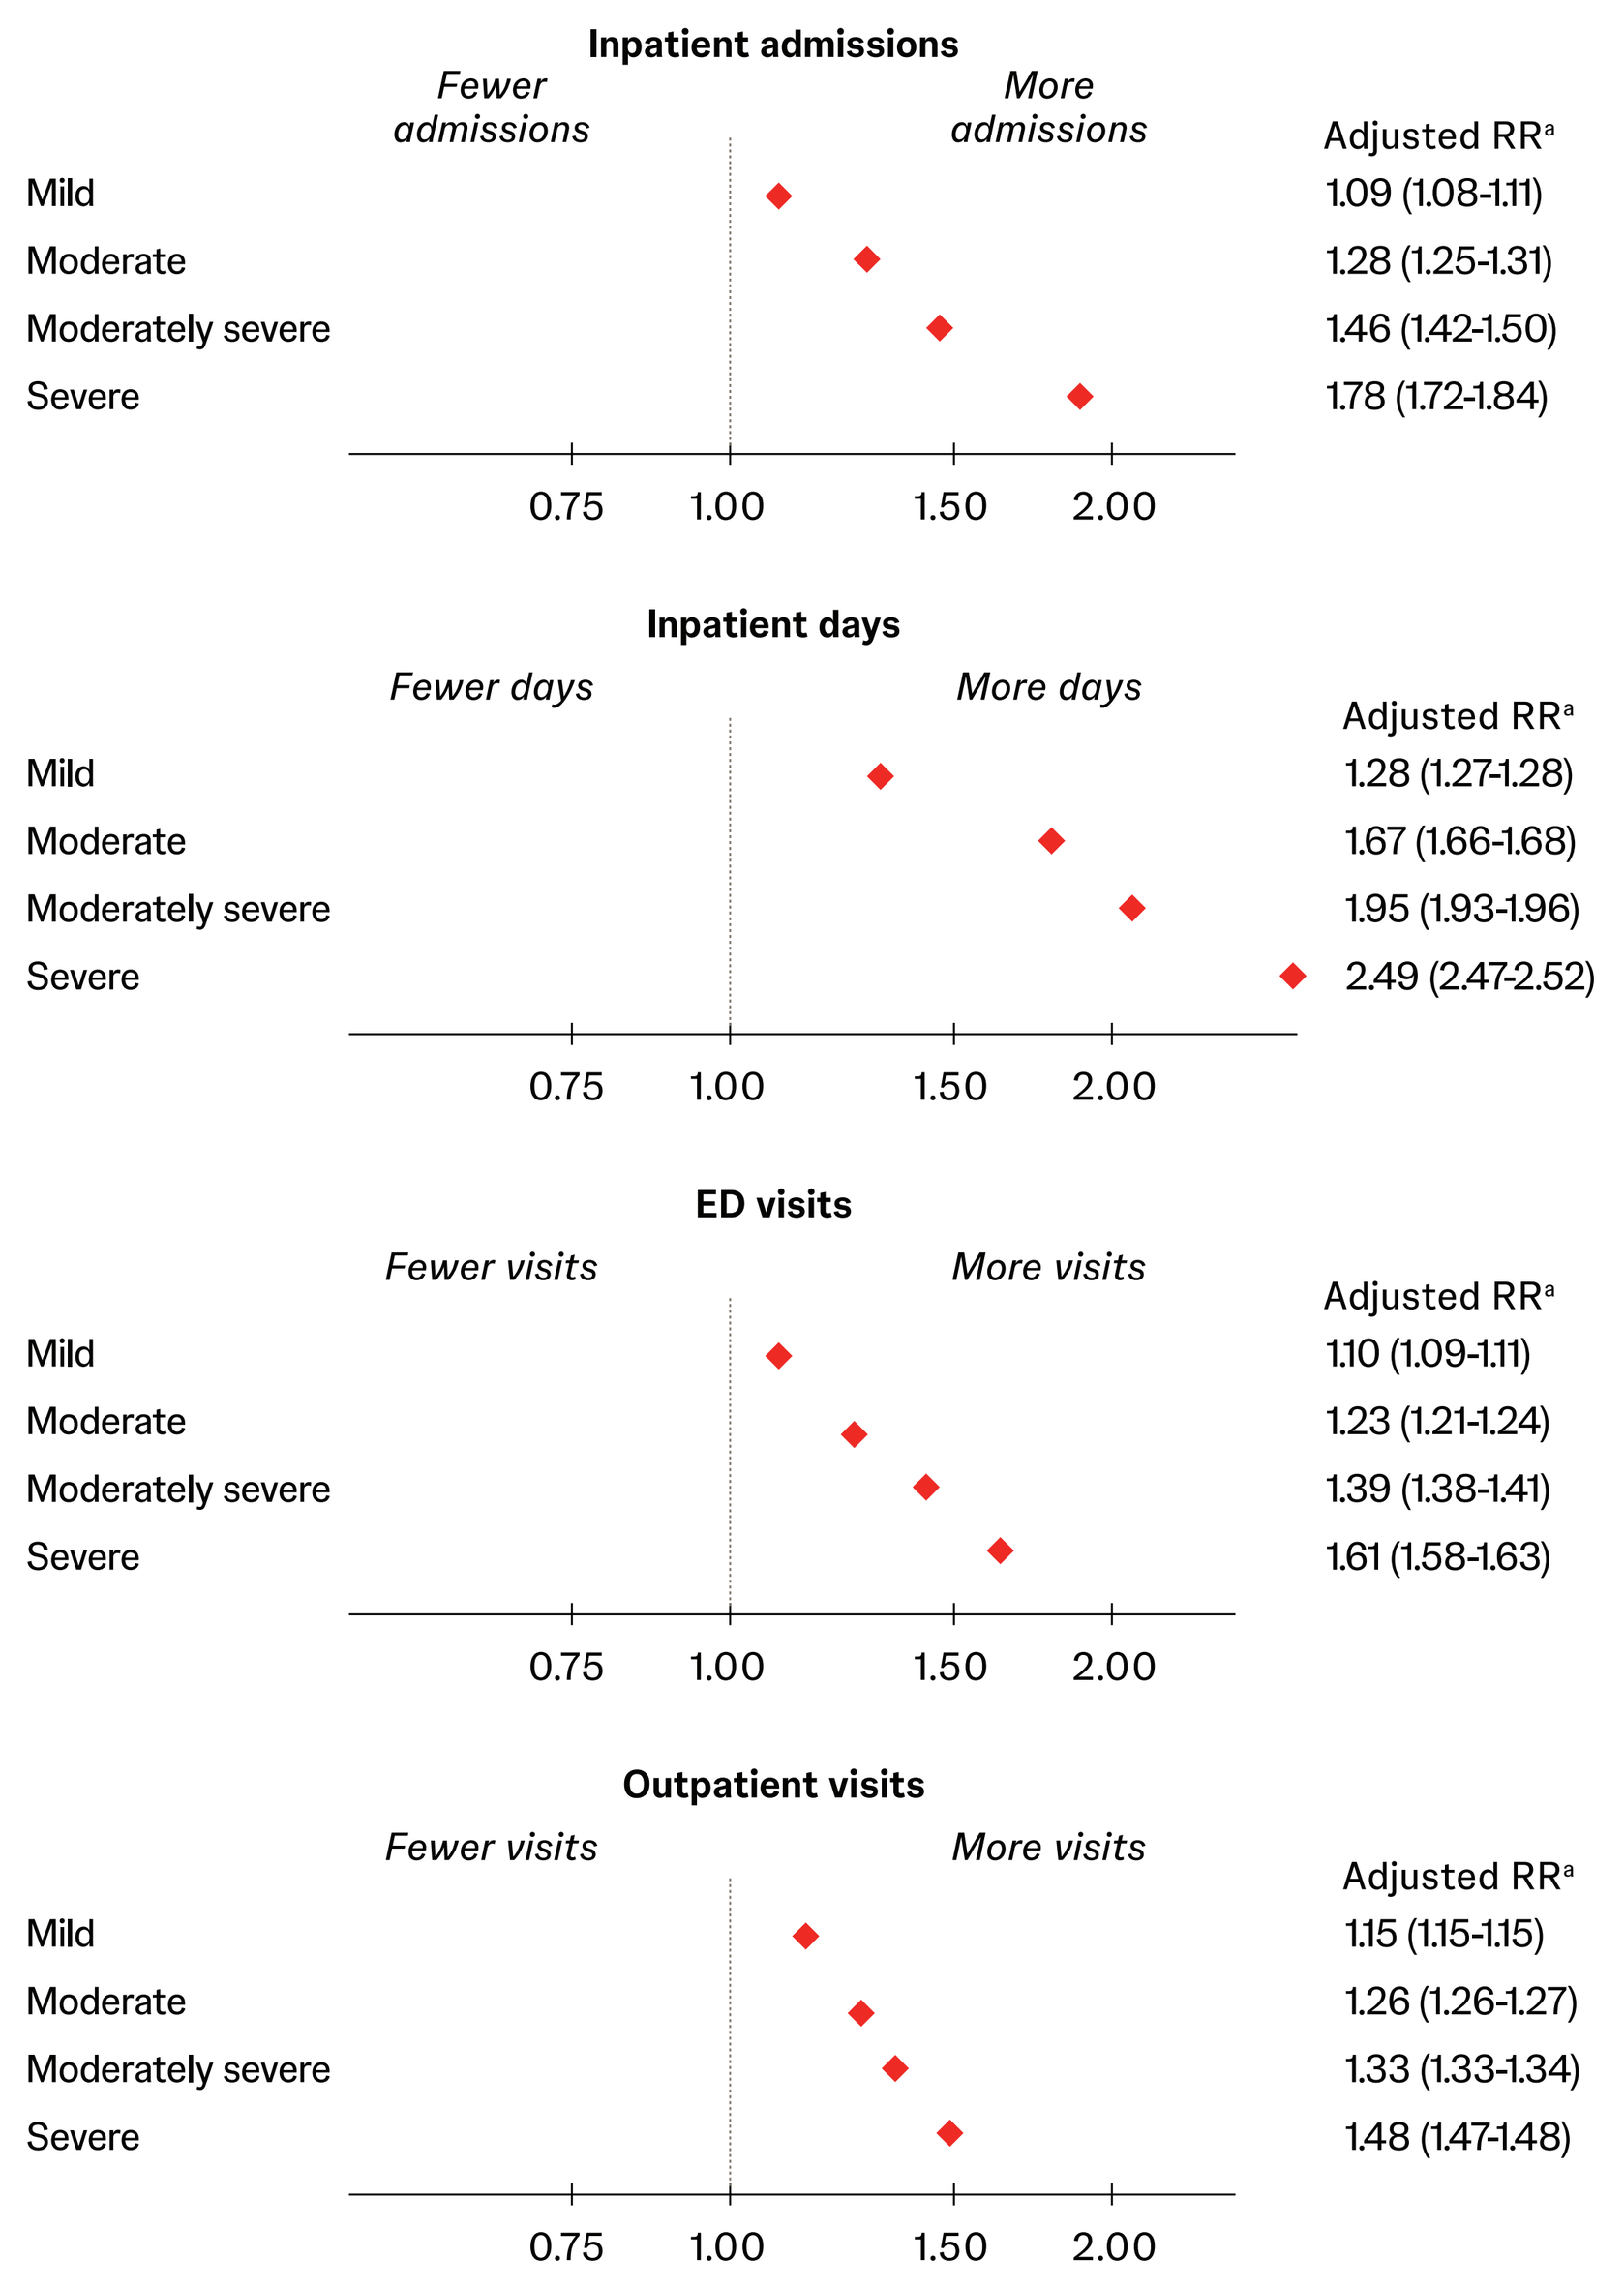
Inpatient admissions
Fewer admissions
More admissions
Adjusted RR[a]
Mild 1.09 (1.08-1.11)
Moderate 1.28 (1.25-1.31)
Moderately severe 1.46 (1.42-1.50)
Severe 1.78 (1.72-1.84)
0.75 1.00 1.50 2.00
Inpatient days
Fewer days
More days
Adjusted RR[a]
Mild 1.28 (1.27-1.28)
Moderate 1.67 (1.66-1.68)
Moderately severe 1.95 (1.93-1.96)
Severe 2.49 (2.47-2.52)
0.75 1.00 1.50 2.00
ED visits
Fewer visits
More visits
Adjusted RR[a]
Mild 1.10 (1.09-1.11)
Moderate 1.23 (1.21-1.24)
Moderately severe 1.39 (1.38-1.41)
Severe 1.61 (1.58-1.63)
0.75 1.00 1.50 2.00
Outpatient visits
Fewer visits
More visits
Adjusted RR[a]
Mild 1.15 (1.15-1.15)
Moderate 1.26 (1.26-1.27)
Moderately severe 1.33 (1.33-1.34)
Severe 1.48 (1.47-1.48)
0.75 1.00 1.50 2.00

ED, emergency department; HRU, healthcare resource utilization; PHQ-9, Patient Health Questionnaire-9; RR, rate ratio.
[a]All comparisons were statistically significant ($P < 0.001$).
The x-axis is displayed on a base-2 logarithmic scale.

Healthcare costs

- Mean annual total healthcare costs were $19,448 for patients with none/minimal depression
 - Relative to none/minimal depression, mean annual total healthcare costs gradually increased by $2,240 to $7,496 across mild to severe depression, respectively **(Figure 3)**
- Each 5-point increase in PHQ-9 score was associated with a $2,048 increase in annual total healthcare costs

Figure 2: Adjusted annual rate ratios of all-cause HRU associated with a 5-point increase in PHQ-9 score[a]

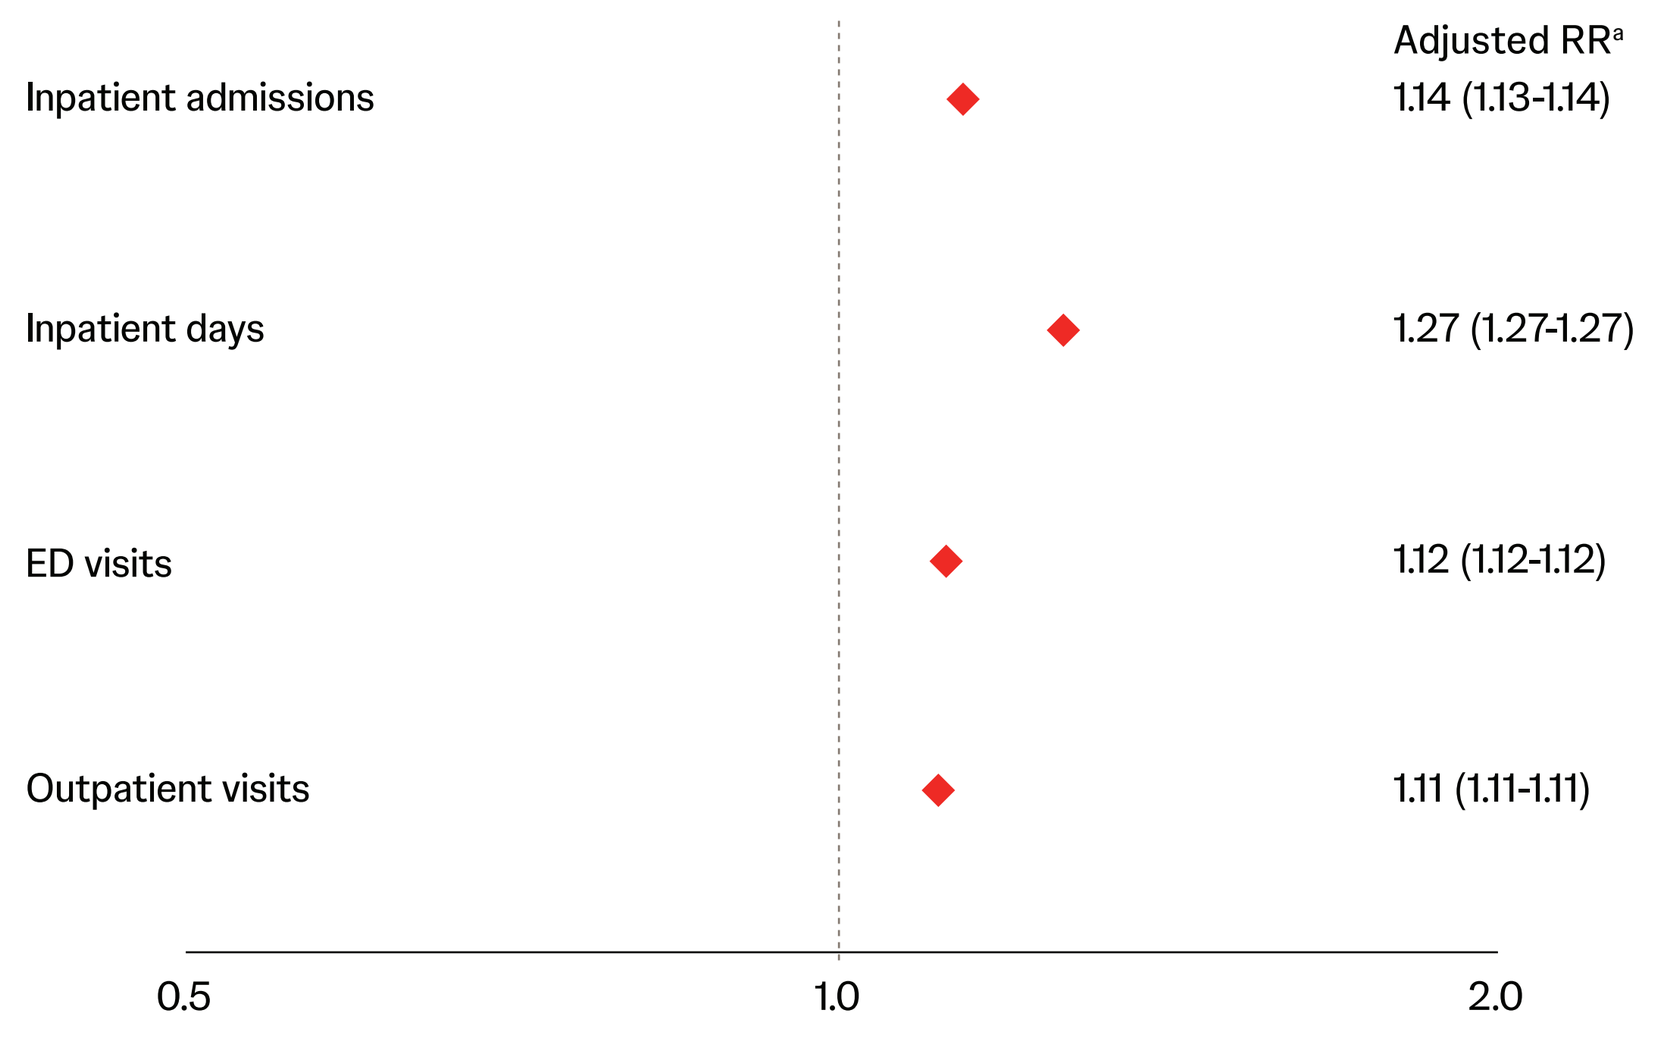
Adjusted RR[a]
Inpatient admissions 1.14 (1.13-1.14)
Inpatient days 1.27 (1.27-1.27)
ED visits 1.12 (1.12-1.12)
Outpatient visits 1.11 (1.11-1.11)
0.5 1.0 2.0

ED, emergency department; HRU, healthcare resource utilization; PHQ-9, Patient Health Questionnaire-9; RR, rate ratio.
[a]All comparisons were statistically significant ($P < 0.001$).
The x-axis is displayed on a base-2 logarithmic scale.

Figure 3: Adjusted mean annual all-cause cost differences by PHQ-9 depression severity relative to "none/minimal" category[a]

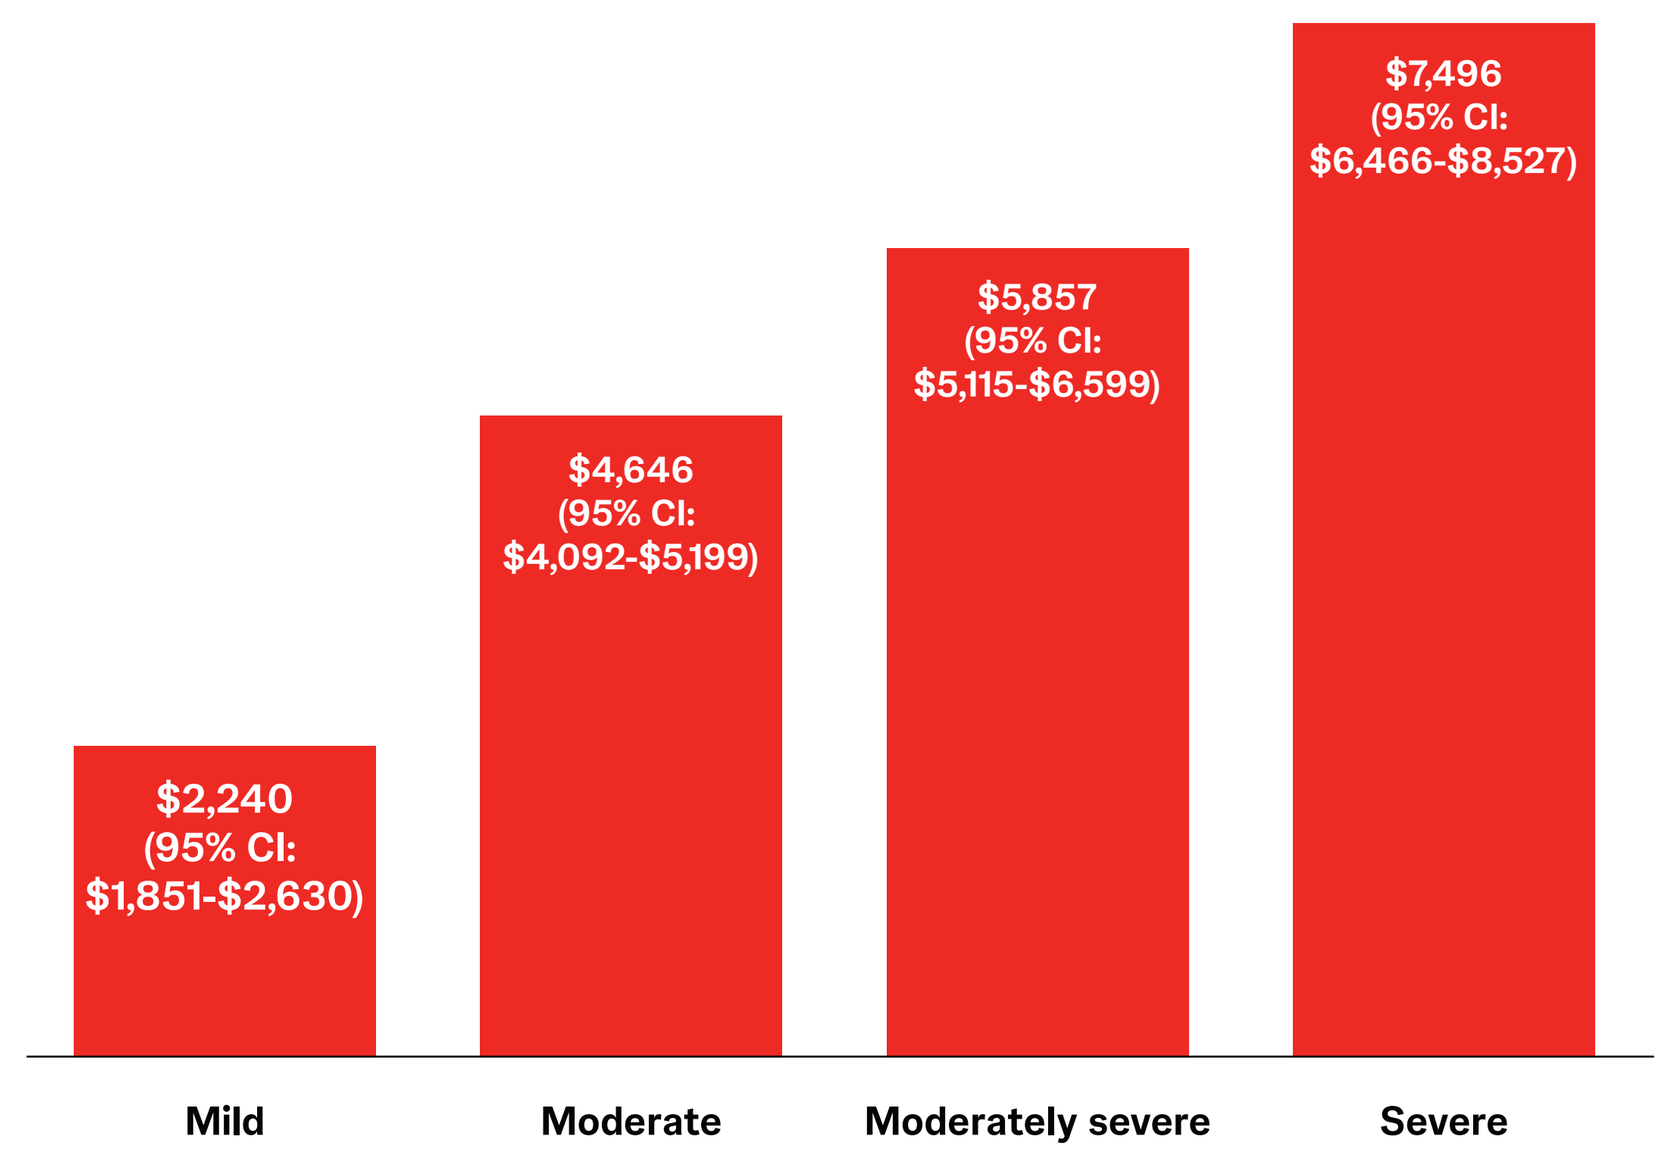
$2,240 (95% CI: $1,851-$2,630)
$4,646 (95% CI: $4,092-$5,199)
$5,857 (95% CI: $5,115-$6,599)
$7,496 (95% CI: $6,466-$8,527)
Mild
Moderate
Moderately severe
Severe

PHQ-9, Patient Health Questionnaire-9.
[a]All comparisons were statistically significant ($P < 0.001$).

Key Takeaways

- Patient age decreased with increasing depression severity, with the youngest patients observed in the severe depression category
- Total healthcare costs and HRU increased with increasing depression severity
- Even a minimally clinically important difference in PHQ-9 score (5 points) was associated with meaningful increases in HRU and healthcare costs

Limitations

- Adjusted analyses may be subject to residual confounding due to unmeasured confounders
- Results may not be generalizable to uninsured or Medicare fee-for-service patients or patients without a documented PHQ-9 score

Conclusions

- Depression severity showed a consistent graded relationship with all-cause HRU and costs across severity categories and with 5-point PHQ-9 increases
- These findings highlight the economic burden of worsening depression and the value of therapies helping patients achieve rapid improvement and remission

Acknowledgements
The authors thank ApotheCom, Yardley, PA, for facilitating poster development

Disclosures
DM and BB are employees of Johnson & Johnson. AV, MZ, DP, AT-S, YW, and PEG are employees of Analysis Group, Inc., a consulting company that has provided paid consulting services to Johnson & Johnson, which funded the development and conduct of this study.

Novel Pathways in Depression

Scan the QR code

The QR code is intended to provide scientific information for individual reference, and the information should not be altered or reproduced in any way.

This study was funded by Johnson & Johnson.

Presented at Psych Congress; September 15-19, 2026; New Orleans, Louisiana, USA

References
1. World Health Organization. Key messages. 2021. Accessed August 31, 2026. https://www.who.int/key-messages. 2. Kroenke K et al. *J Gen Intern Med*. 2001;16(9):606-613.

This material is distributed for scientific purposes by Johnson & Johnson Innovative Medicine and is not for promotional use.