Esketamine Nasal Spray on Anhedonia and Functional Outcomes: Findings from the Ventura Real-World Evidence Study

AUTHORS: Andrew J Cutler,[1] Rakesh Jain,[2] Katharine Henry,[3] Dana Drzayich Jankus,[4] Yordan Godinov,[5] Filip Delaei,[6] Onur Tankaya,[4] Katalin Pungor,[4] Dong-Jing Fu[7]

AFFILIATIONS: [1]SUNY Upstate Medical University, Lakewood Ranch, FL, USA; [2]Texas Tech University School of Medicine, Midland, TX, USA; [3]Johnson & Johnson, Cambridge, MA, USA; [4]Johnson & Johnson, Raritan, NJ, USA; [5]Johnson & Johnson, Sofia, Bulgaria; [6]Johnson & Johnson, Beerse, Belgium; [7]Johnson & Johnson, Titusville, NJ, USA

This material is distributed for scientific purposes by Johnson & Johnson Innovative Medicine and is not for promotional use.

Background

- Anhedonia is a core symptom of major depressive disorder (MDD) associated with poor prognosis and functional impairment[1]
- Previous studies suggest that improvement in anhedonia may be linked to enhanced overall functioning[2]
- Esketamine nasal spray (NS) is approved for treatment-resistant depression as monotherapy or in conjunction with an oral antidepressant (OAD) and for depressive symptoms in patients with MDD and acute suicidal ideation or behavior in conjunction with an OAD[3]
- Esketamine NS + OAD showed decrease in anhedonia (patient- and physician-reported)[4]
- Post-hoc analysis from 2 phase 3 studies showed improvements in anhedonia symptoms in short-term (TRANSFORM-2) and long-term (SUSTAIN-2) treatment[5]

Methods

- Ventura-real-world evidence (RWE) (NCT05841030) was a prospective, noninterventional, 12-month study to document disease characteristics and outcomes in real-world standard of care (SOC) use of available treatments among patients with MDD with anhedonia
- Patients enrolled had inadequate response to currently ongoing antidepressant treatment, inclusive of an selective serotonin reuptake inhibitor (SSRI) /serotonin-norepinephrine reuptake inhibitor (SNRI) and initiated physicians' choice of adjunctive treatment for MDD, including esketamine NS, based on prescribing information
- Outcome measures were assessed at baseline, Week 6, Month 6 and Month 12[6]
 - Depression severity (Montgomery-Åsberg Depression Rating Scale [MADRS], Patient Health Questionnaire-9 item [PHQ-9])
 - Anhedonia (Snaith-Hamilton Pleasure Scale [SHAPS], Dimensional Anhedonia Rating Scale [DARS], MADRS 8, MADRS anhedonia subscale, Patient Global Impression Scale [PGI-S] anhedonia)
 - Functional impairment (Work Productivity and Activity Impairment [WPAI])
 - Health-related quality of life (HRQoL) (EuroQoL-5-dimension-5-level [EQ-5D-5L], EQ-5D Visual Analogue Scale [VAS])
- The presented post-hoc analysis included all patients who initiated esketamine NS at baseline, using an intention-to-treat (ITT) assumption
- Descriptive and Spearman correlation analyses were performed
- A sensitivity analysis was conducted using patients "as treated" with esketamine NS, using only data from patients while they were on esketamine NS during the study

DARS, Dimensional Anhedonia Rating Scale; EQ-5D-5L, EuroQoL-5-dimension-5-level; HRQoL, health-related quality of life; ITT, intention-to-treat; MADRS, Montgomery-Åsberg Depression Rating Scale; NS, nasal spray; OAD, oral antidepressant; PGI-S, Patient Global Impression Scale; PHQ-9, Patient Health Questionnaire-9 item; RWE, real-world evidence; SHAPS, Snaith-Hamilton Pleasure Scale; SNRI, serotonin-norepinephrine reuptake inhibitor; SOC, standard of care; SSRI, selective serotonin reuptake inhibitor; VAS, Visual Analogue Scale; WPAI, Work Productivity and Activity Impairment.

Results

- Patient dispostion of esketamine NS is shown in **Figure 1**.
- Patients had moderate to severe depression and a high level of anhedonia based on SHAPS and DARS (**Table 1**)

FIGURE 1: Patient Disposition

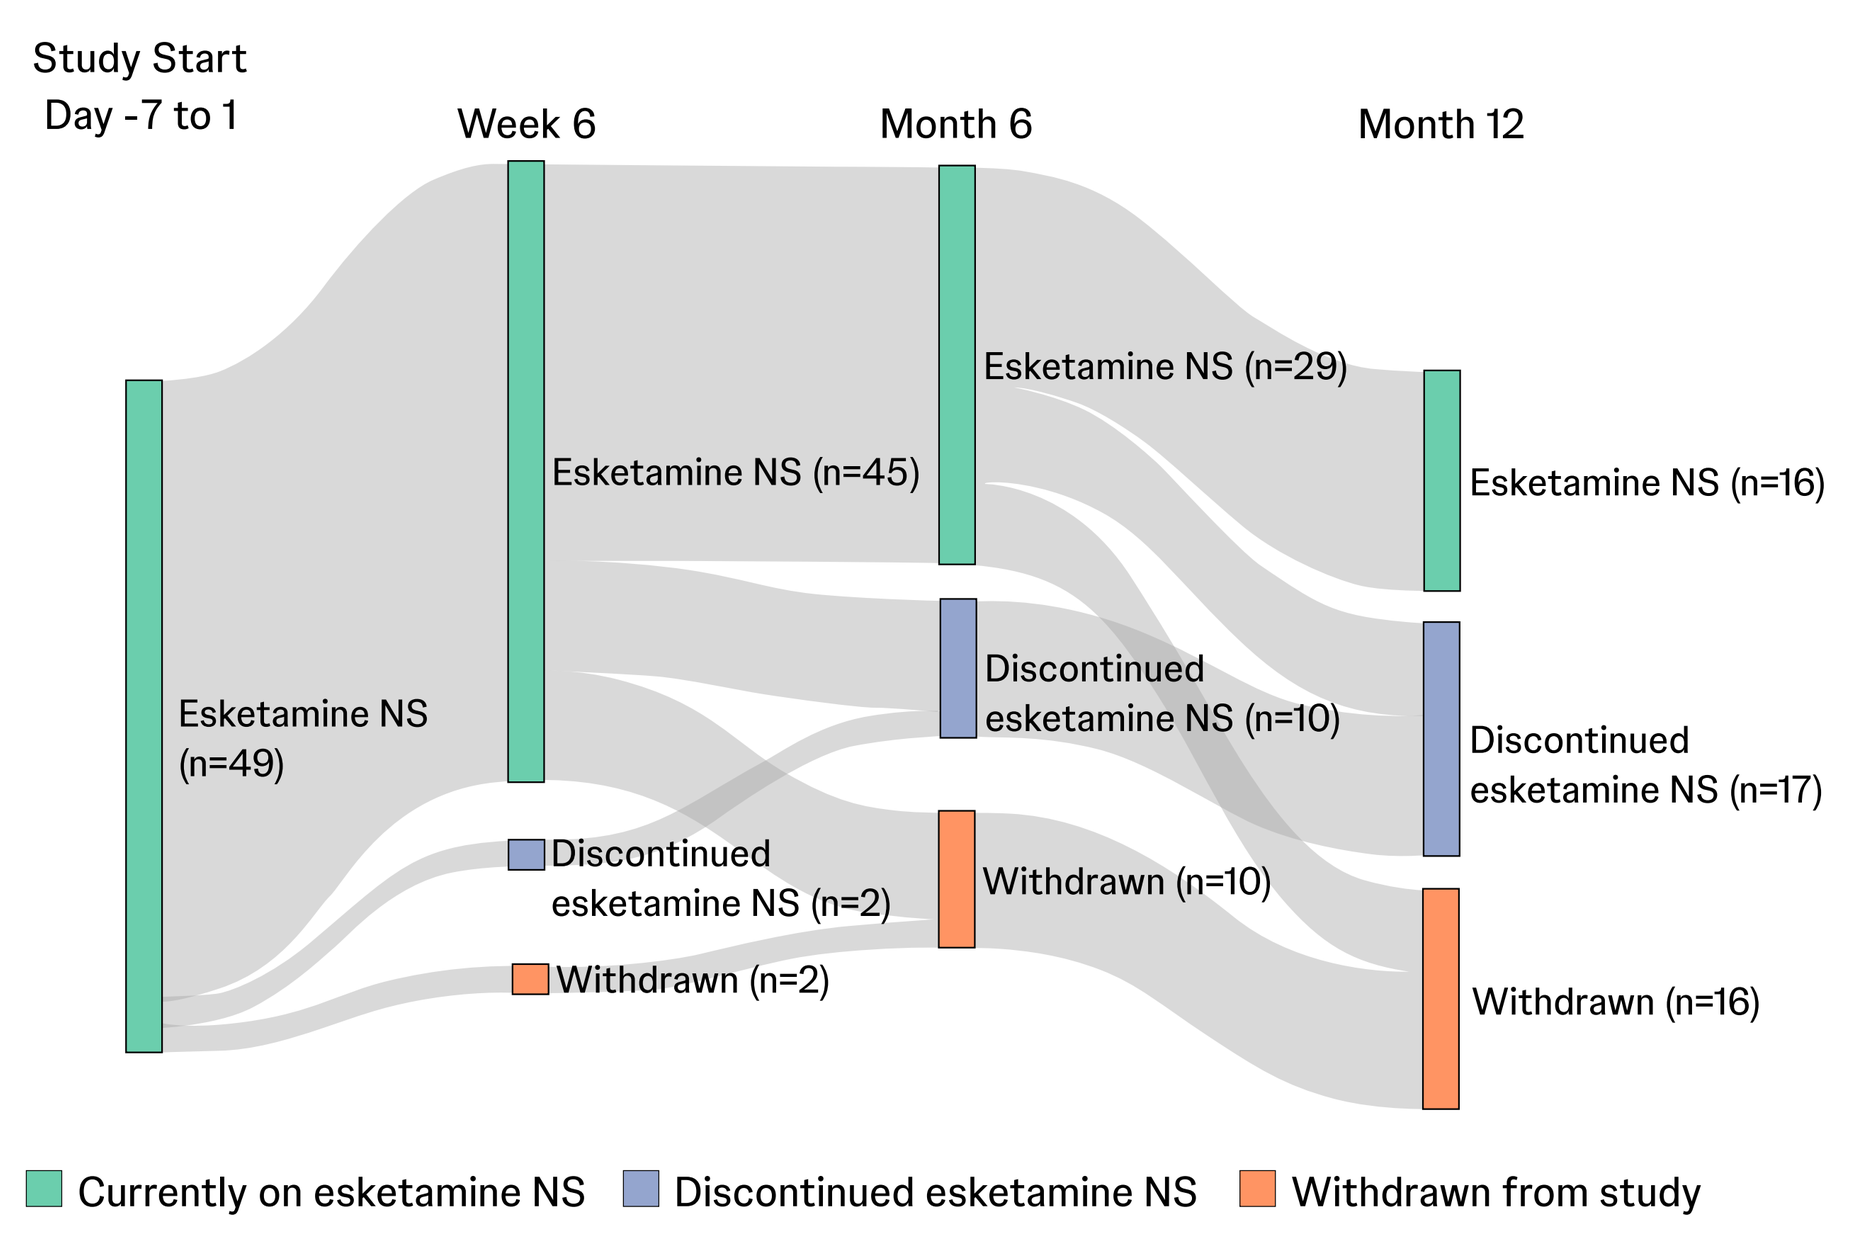
Study Start
Day -7 to 1
Week 6
Month 6
Month 12
Esketamine NS (n=49)
Esketamine NS (n=45)
Discontinued esketamine NS (n=2)
Withdrawn (n=2)
Esketamine NS (n=29)
Discontinued esketamine NS (n=10)
Withdrawn (n=10)
Esketamine NS (n=16)
Discontinued esketamine NS (n=17)
Withdrawn (n=16)
Currently on esketamine NS
Discontinued esketamine NS
Withdrawn from study

TABLE 1: Patient Characteristics for Esketamine NS + OAD (N=49)

Baseline Characteristics	Median (IQR)	Mean (SD)
Age, years	54 (44–62)	51.6 (13.2)
Female/male, n (%)	30/19 (61%/39%)	
MADRS	30 (27–34)	30.5 (5.5)
PHQ-9	18 (13–23)	17.6 (6.0)
SHAPS	37 (34–40)	36.9 (5.5)
DARS	24 (16–35)	25.1 (11.9)
MADRS anhedonia subscale	19 (16–21)	18.6 (3.9)
MADRS Q8	4 (3–4)	3.7 (1.0)
PHQ-9 Q1	3 (2–4)	2.5 (0.7)
Age MDD diagnosis, years	26 (18–47)	29.5 (14.6)
Duration of current episode, weeks	43 (20–141)	184.3 (429.5)
Total MDD episodes	3 (2–7)	5.4 (5.5)

DARS, Dimensional Anhedonia Rating Scale; IQR, interquartile range; MADRS, Montgomery-Åsberg Depression Rating Scale; MDD, major depressive disorder; NS, nasal spray; OAD, oral antidepressant; PHQ-9, Patient Health Questionnaire; SHAPS, Snaith-Hamilton Pleasure Scale; SD, standard deviation.

Esketamine NS + OAD was Associated with Improvements in Anhedonia Severity and Overall MDD Symptoms

- Patients treated with esketamine NS + OAD showed continued improvement in MDD severity and in anhedonia symptoms (**Figure 2A**)
- Improvement in anhedonia severity was demonstrated in changes in anhedonia specific scales and relevant depression scales (**Figure 2**)

FIGURE 2: MDD and Anhedonia Measures

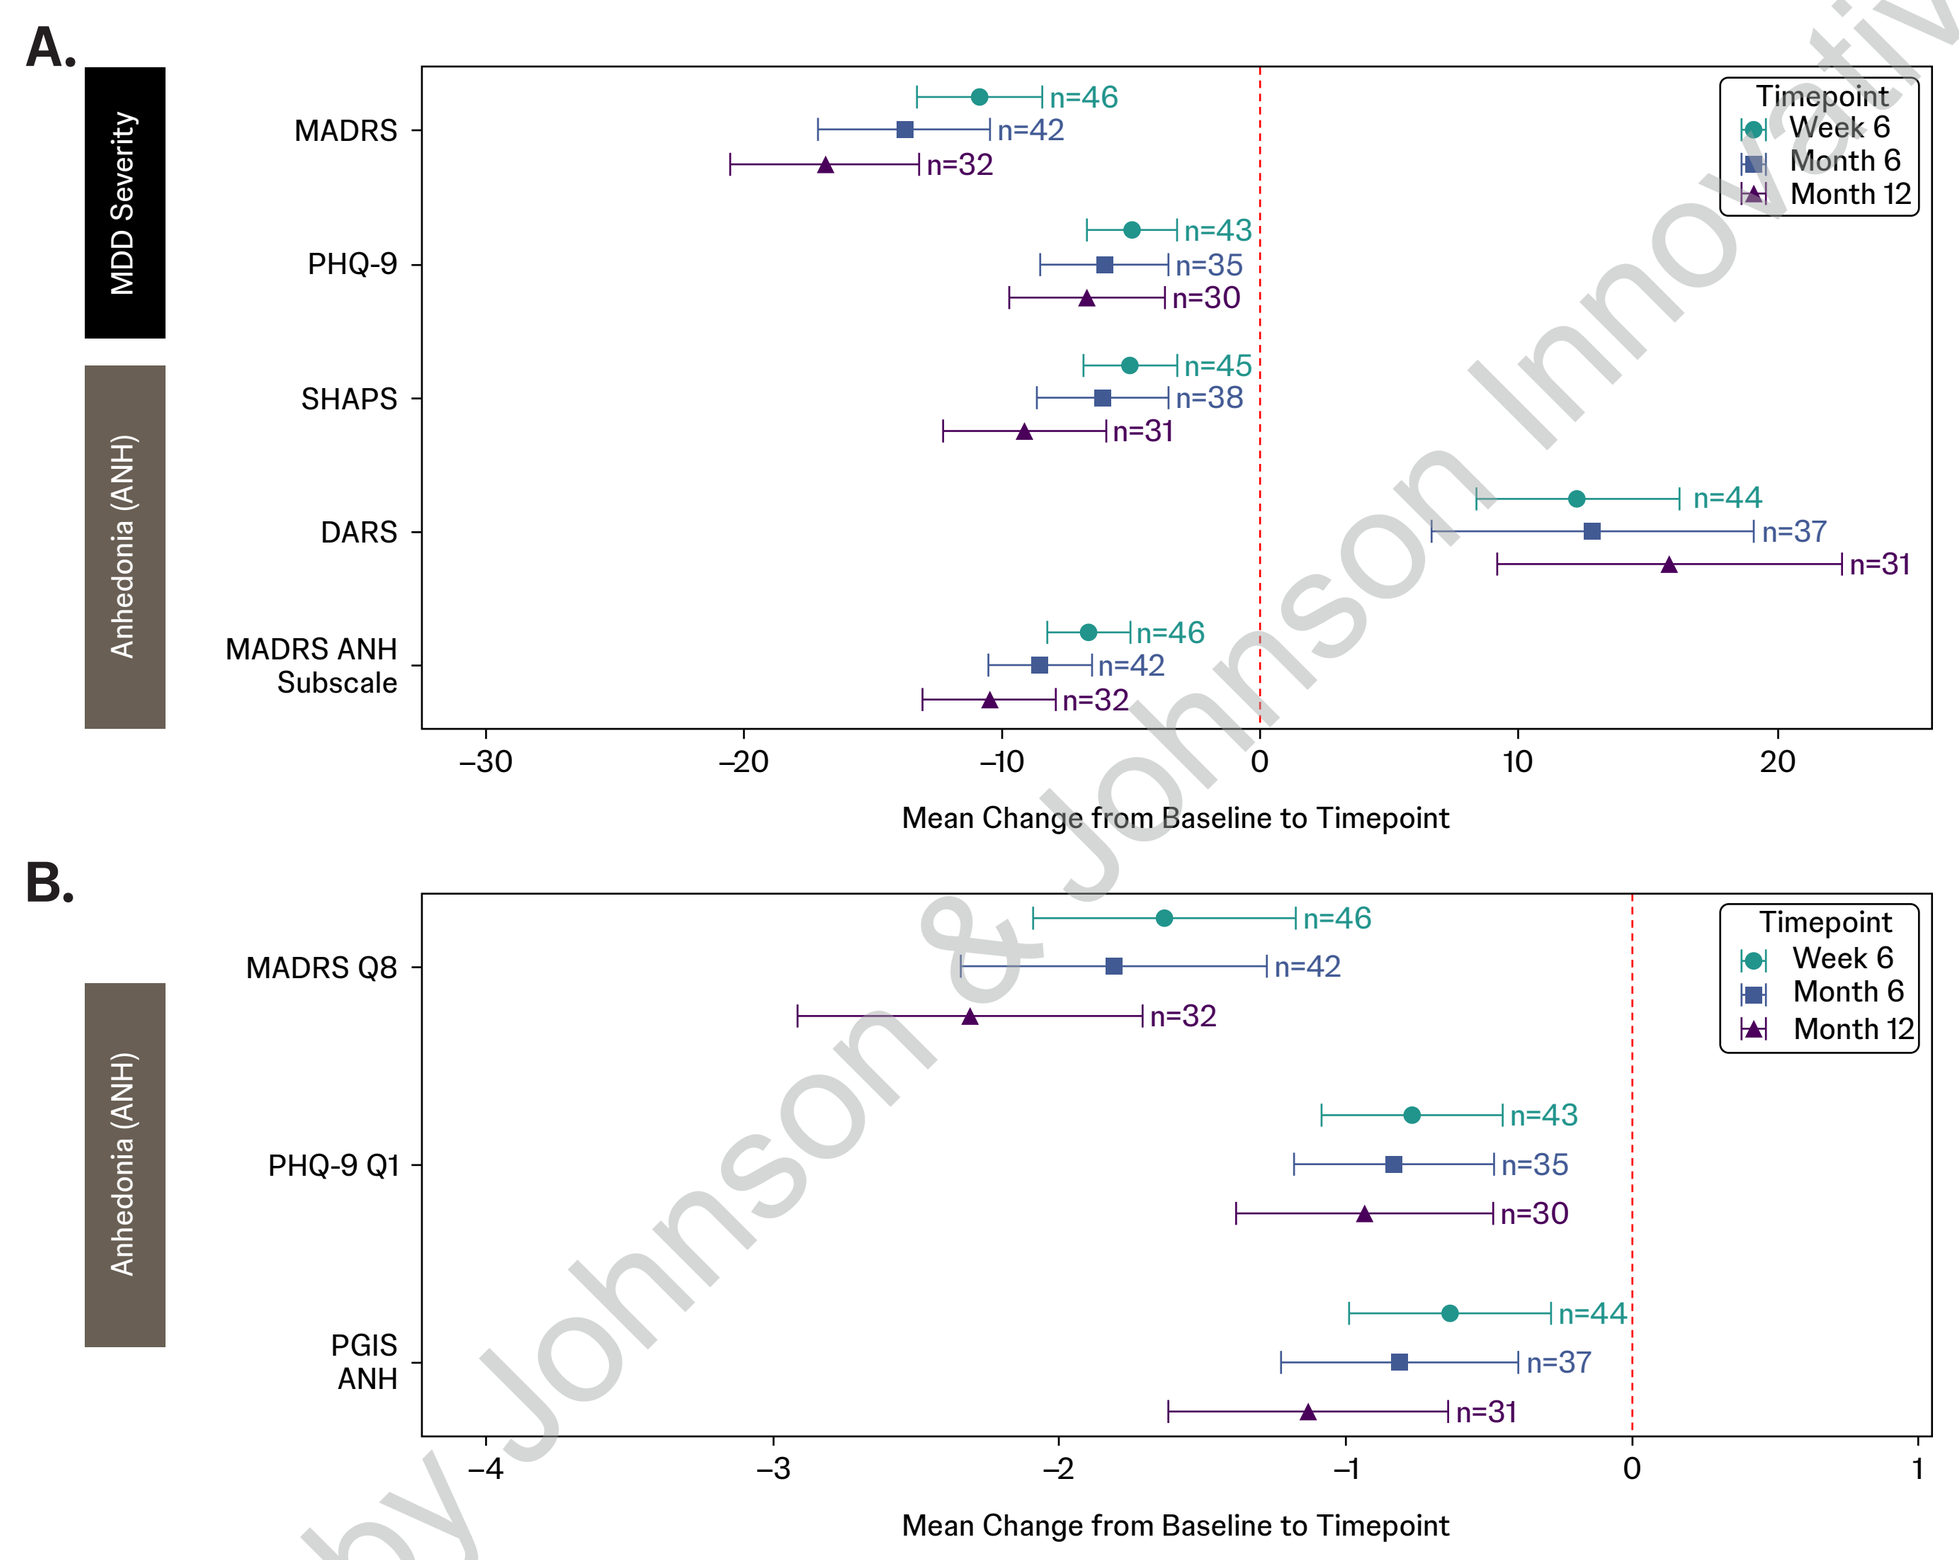
A.
MDD Severity
Anhedonia (ANH)
MADRS
PHQ-9
SHAPS
DARS
MADRS ANH Subscale
Timepoint
Week 6
Month 6
Month 12
Mean Change from Baseline to Timepoint
B.
MADRS Q8
PHQ-9 Q1
PGIS ANH
Mean Change from Baseline to Timepoint

Montgomery-Åsberg Depression Rating Scale (MADRS): total score 0-60
Patient Health Questionnaire (PHQ)-9: total score 0-27
Snaith-Hamilton Pleasure Scale (SHAPS): total score 14-56
Dimensional Anhedonia Rating Scale (DARS): total score 0-68
Patient Global Impression Scale (PGI-S) ANH (anhedonia): score 1-5
(Note: DARS has opposite directionality as other scores, with higher values indicating lower severity)

Esketamine NS + OAD was Associated with Functional Improvements

- EQ-5D-5L domains of usual activities and anxiety/depression showed improvements (**Figure 3**)
- EQ-5D VAS scores showed improvement

FIGURE 3: HRQoL Improvement Measures

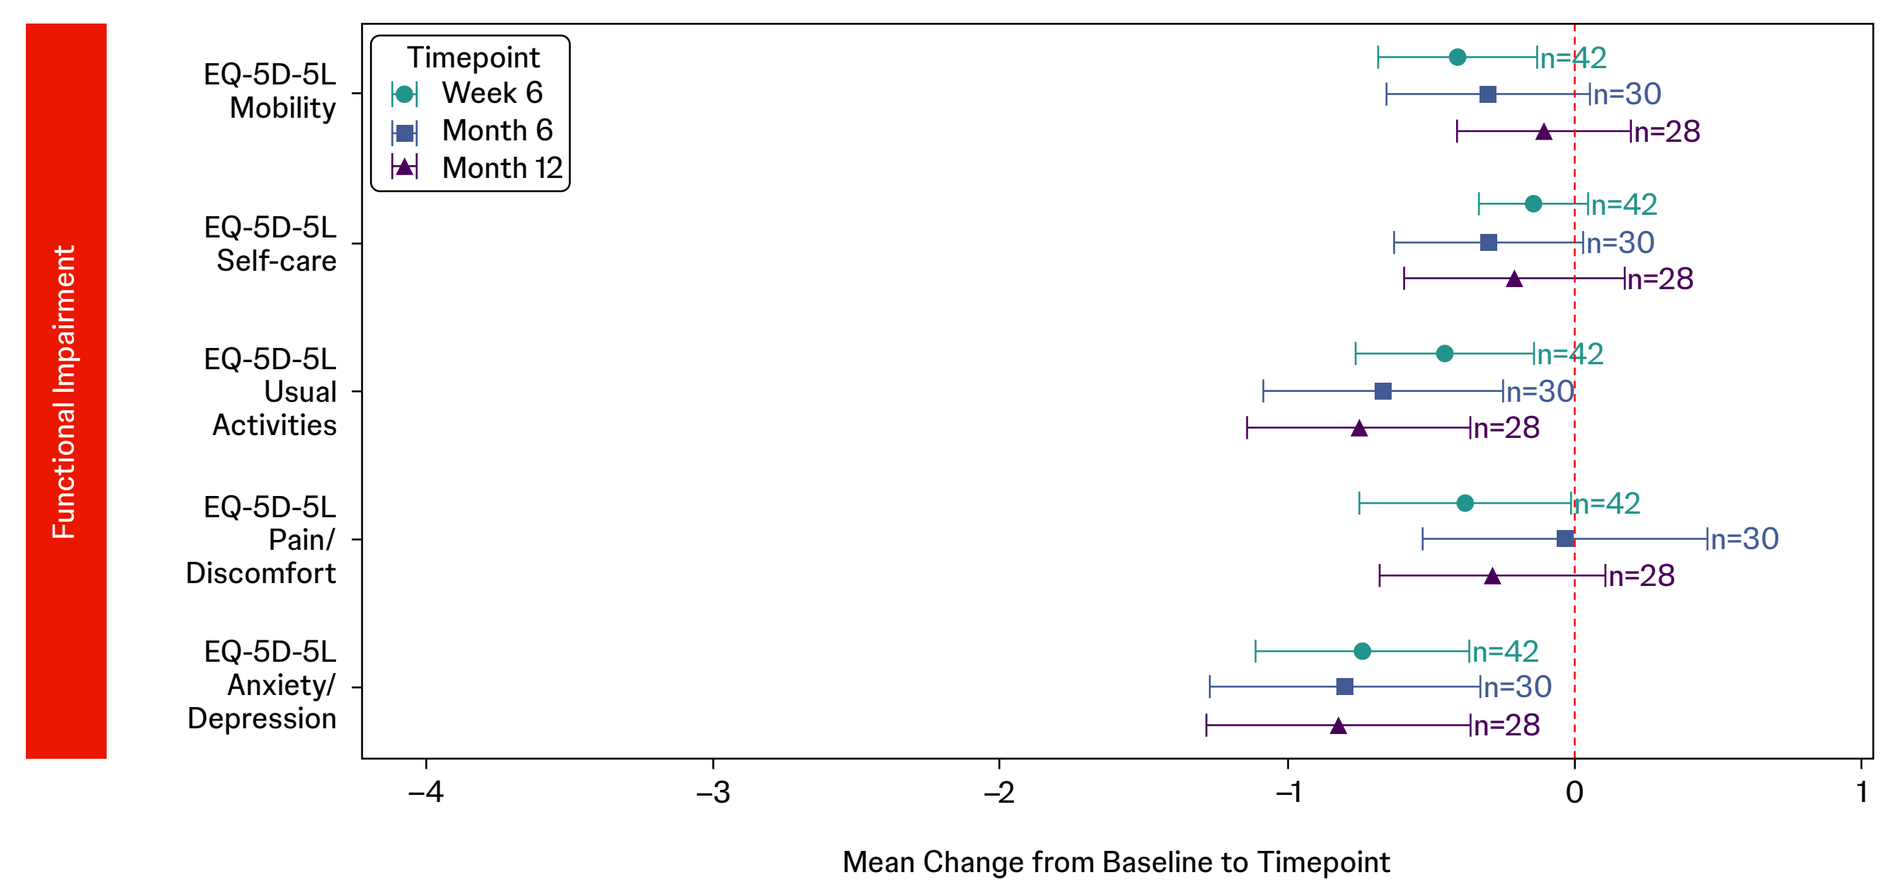
Functional Impairment
EQ-5D-5L Mobility
EQ-5D-5L Self-care
EQ-5D-5L Usual Activities
EQ-5D-5L Pain/ Discomfort
EQ-5D-5L Anxiety/ Depression
Timepoint
Week 6
Month 6
Month 12
Mean Change from Baseline to Timepoint

EQ-5D-5L, EuroQoL-5-dimension-5-level; HRQoL, health-related quality of life.

- Esketamine NS + OAD showed improvements in WPAI categories (**Figure 4**)
- Limited sample size in the esketamine NS-started patients with WPAI work-related responses limit ability to evaluate any associations between esketamine NS + OAD use and WPAI work-related functional changes, especially at later timepoints

FIGURE 4: Work Productivity Measures

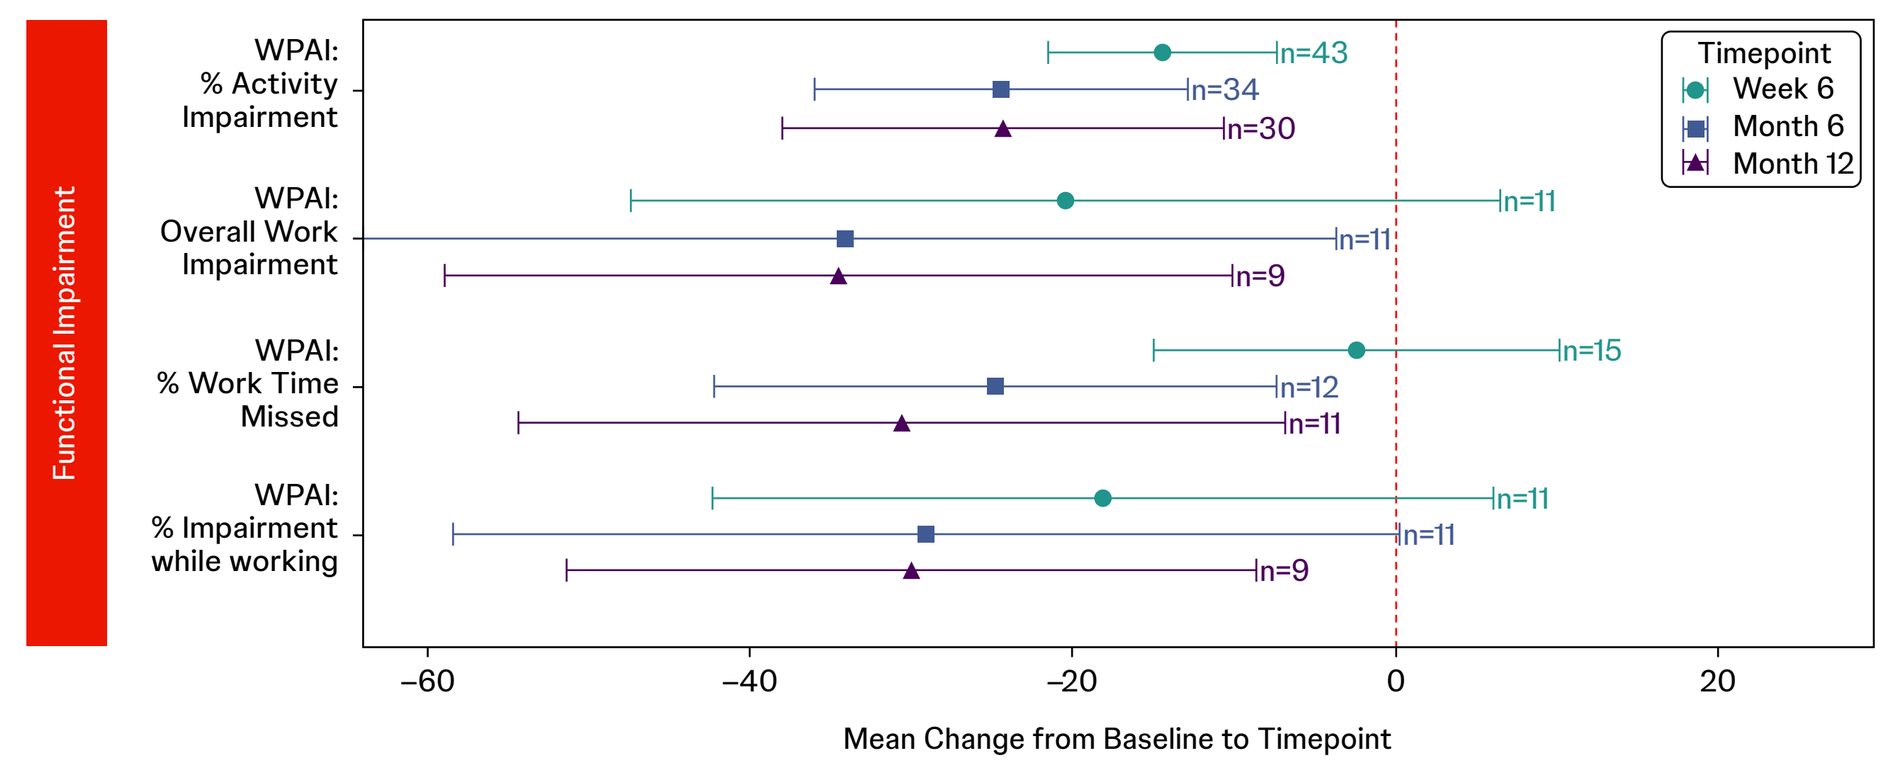
Functional Impairment
WPAI: % Activity Impairment
WPAI: Overall Work Impairment
WPAI: % Work Time Missed
WPAI: % Impairment while working
Timepoint
Week 6
Month 6
Month 12
Mean Change from Baseline to Timepoint

HRQoL, health-related quality of life; WPAI, Work Productivity and Activity Impairment

Changes in Anhedonia Symptom Severity Strongly Correlated with Changes in MDD Severity and Functional Impairment

- Moderate correlation between changes in SHAPS/DARS and changes in other measures of anhedonia at Week 6 and strong correlation at Month 6 and Month 12 were observed (**Figure 5A/B**)
- Correlations between changes in SHAPS and MDD severity scales strengthened from Week 6 to Month 6 (**Figure 5A**)
- Correlations between changes in DARS were similar to SHAPS observations (**Figure 5B**)

Note: DARS is scaled in opposite direction (higher DARS is less severe)

FIGURE 5: Spearman Correlation Between Change in Scales from Baseline to Timepoint A) SHAPS; B) DARS

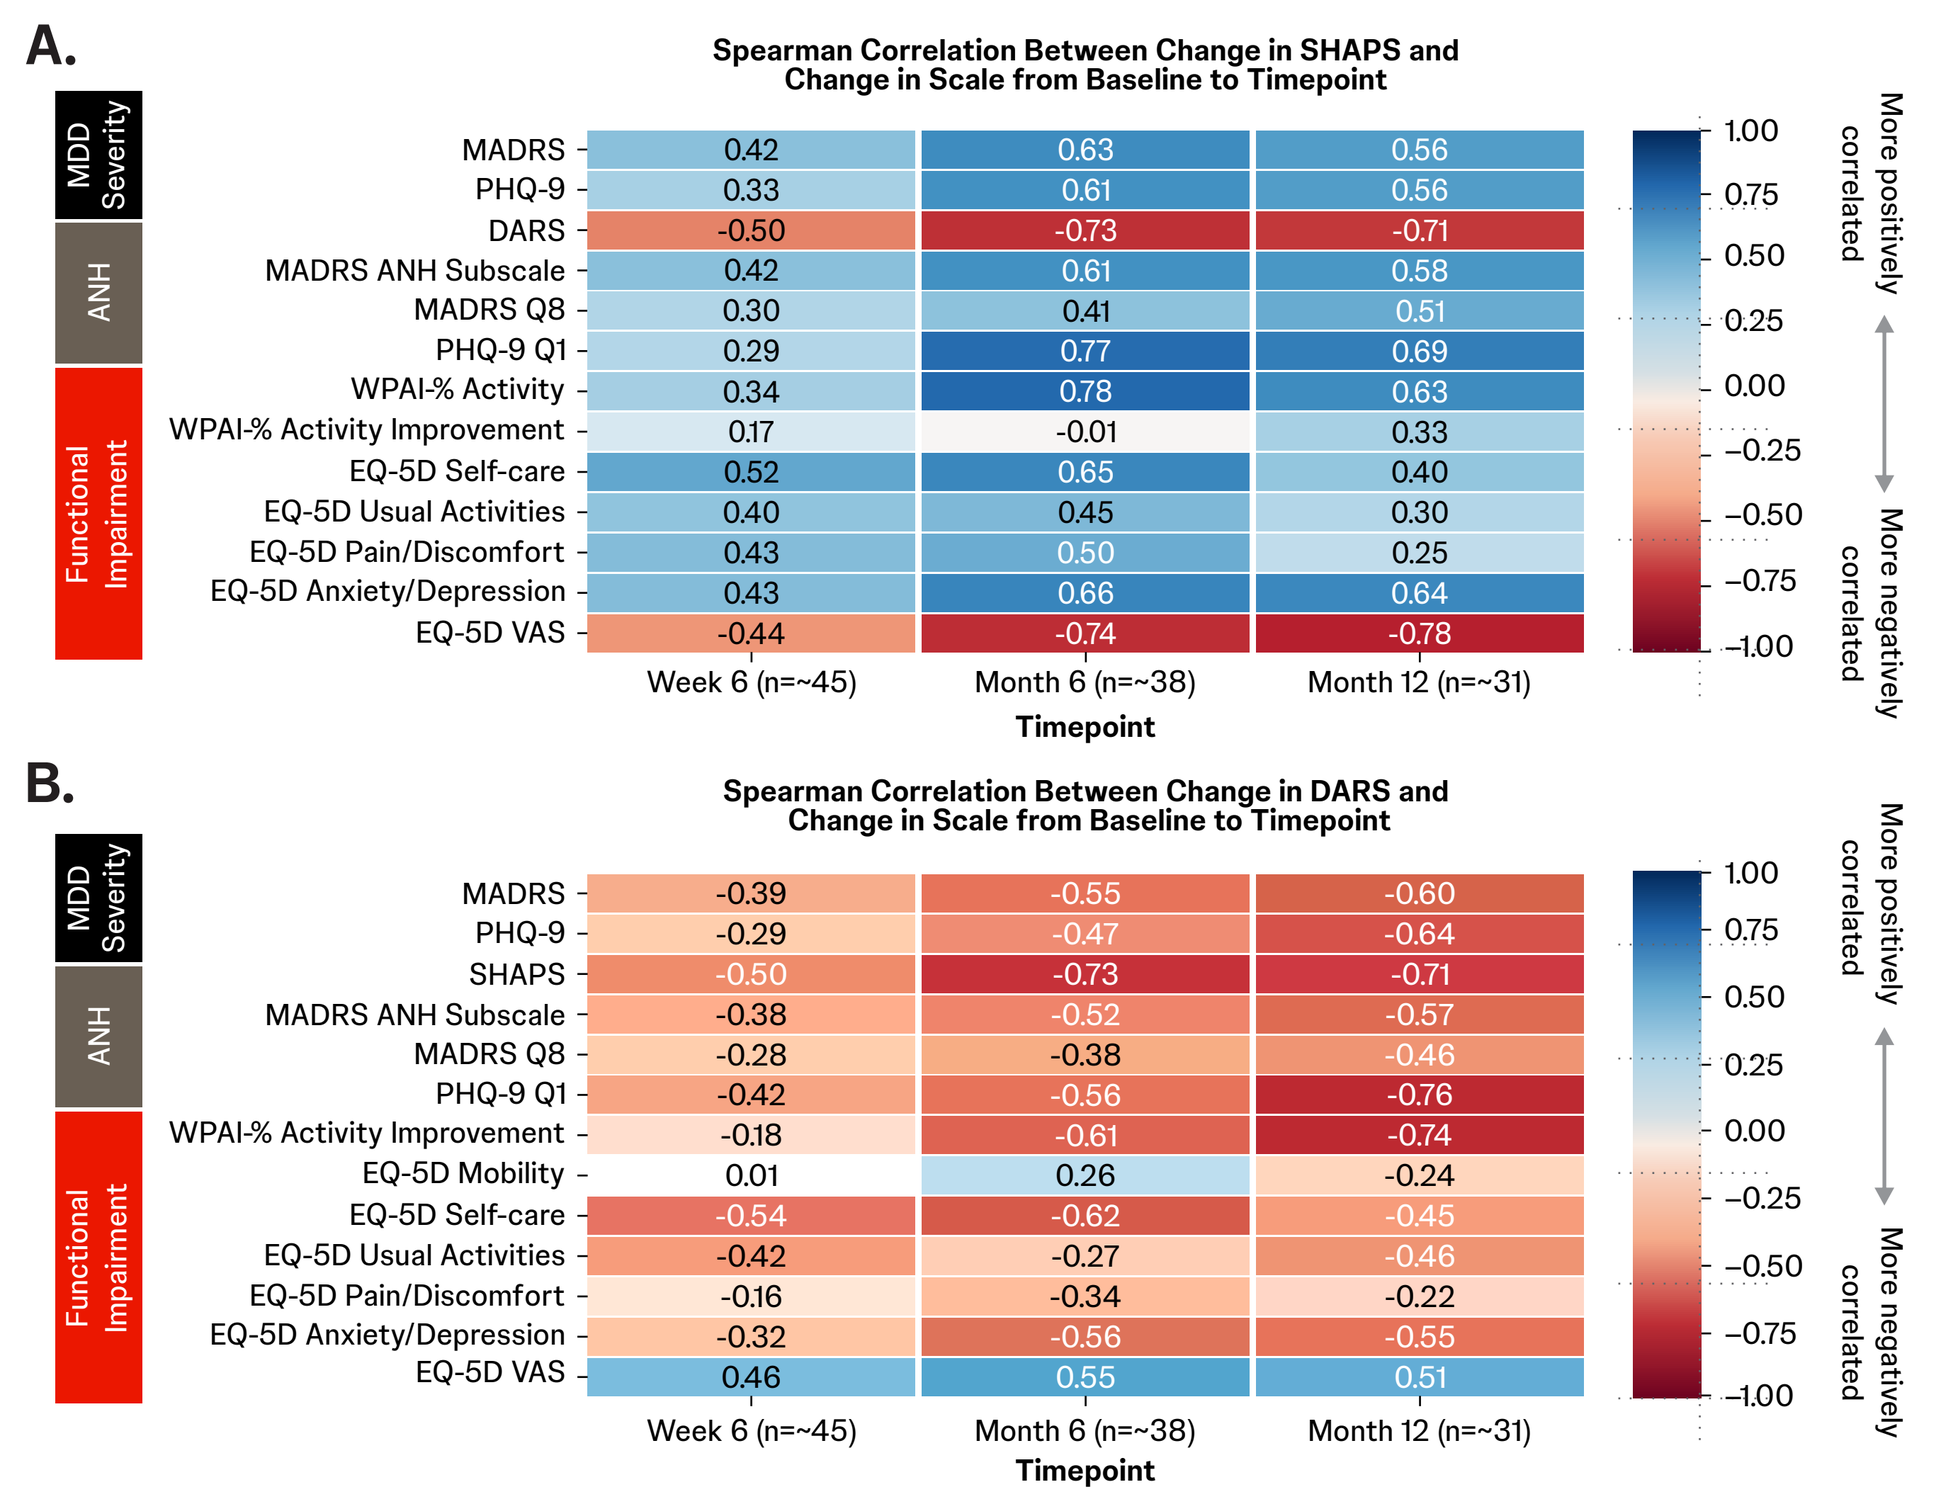
A.
Spearman Correlation Between Change in SHAPS and Change in Scale from Baseline to Timepoint
MDD Severity
ANH
Functional Impairment
MADRS 0.42 0.63 0.56
PHQ-9 0.33 0.61 0.56
DARS -0.50 -0.73 -0.71
MADRS ANH Subscale 0.42 0.61 0.58
MADRS Q8 0.30 0.41 0.51
PHQ-9 Q1 0.29 0.77 0.69
WPAI-% Activity 0.34 0.78 0.63
WPAI-% Activity Improvement 0.17 -0.01 0.33
EQ-5D Self-care 0.52 0.65 0.40
EQ-5D Usual Activities 0.40 0.45 0.30
EQ-5D Pain/Discomfort 0.43 0.50 0.25
EQ-5D Anxiety/Depression 0.43 0.66 0.64
EQ-5D VAS -0.44 -0.74 -0.78
Week 6 (n=~45)
Month 6 (n=~38)
Month 12 (n=~31)
Timepoint
More positively correlated
More negatively correlated
B.
Spearman Correlation Between Change in DARS and Change in Scale from Baseline to Timepoint
MADRS -0.39 -0.55 -0.60
PHQ-9 -0.29 -0.47 -0.64
SHAPS -0.50 -0.73 -0.71
MADRS ANH Subscale -0.38 -0.52 -0.57
MADRS Q8 -0.28 -0.38 -0.46
PHQ-9 Q1 -0.42 -0.56 -0.76
WPAI-% Activity Improvement -0.18 -0.61 -0.74
EQ-5D Mobility 0.01 0.26 -0.24
EQ-5D Self-care -0.54 -0.62 -0.45
EQ-5D Usual Activities -0.42 -0.27 -0.46
EQ-5D Pain/Discomfort -0.16 -0.34 -0.22
EQ-5D Anxiety/Depression -0.32 -0.56 -0.55
EQ-5D VAS 0.46 0.55 0.51
Week 6 (n=~45)
Month 6 (n=~38)
Month 12 (n=~31)
Timepoint

ANH, anhedonia; DARS, Dimensional Anhedonia Rating Scale; EQ-5D, EuroQoL-5-dimension; MADRS, Montgomery-Åsberg Depression Rating Scale; MDD, major depressive disorder; PHQ-9, Patient Health Questionnaire; SHAPS, Snaith-Hamilton Pleasure Scale; WPAI, Work Productivity and Activity Impairment; VAS, Visual Analogue Scale

Note: white indicates no correlation, color intensity indicates strength of correlation (darker color = stronger correlation).

74

Key takeaways

Using real-world findings from the Ventura-RWE prospective, observational study, esketamine NS + OAD demonstrated improvements in anhedonia measures and gains in functional outcomes.

Targeting anhedonia remains an important treatment goal in MDD patients with inadequate response to previous antidepressants as it is strongly correlated with overall depression symptom improvement.

Conclusions

Ventura-RWE included a small cohort of patients treated with esketamine NS + OAD who completed comprehensive assessments of depression and anhedonia severity.

Esketamine NS + OAD was associated with improvements in anhedonia and functional impairment, as well as overall MDD severity.

- Improvement in anhedonia was correlated to improvement in functioning and multiple HRQoL measures.

Acknowledgements

This presentation was sponsored by Johnson & Johnson. Medical writing support was provided by Denise Balog, PharmD (inSeption Group), funded by Johnson & Johnson and Anthony Gaba, PhD (Johnson & Johnson) under the direction of the authors in accordance with Good Publication Practice guidelines (*Ann Intern Med.* 2022;175:1298–1304). Poster layout was performed by Sandeep Chavan (SIRO Clinpharm Pvt. Ltd.), funded by Johnson & Johnson.

Disclosures

Andrew J Cutler is a consultant and/or on an advisory board for AbbVie, Acadia, Actinogen, Alfasigma, Alkermes, Anavex Life Sciences, Arrivo BioVentures, Autobahn Therapeutics, Axsome, Aytu Biopharma, Bristol Myers Squibb, Collegium Pharmaceutical, Definium Therapeutics, 4M Therapeutics, Helus Pharma, Incannex Healthcare, Intra-Cellular Therapies, J&J Innovative Medicine, Jazz Pharma, Knight Therapeutics, Kye Pharmaceuticals, Kuvatris Therapeutics, LivaNova, Lundbeck, Luye Pharma, MapLight Therapeutics, Mentavi, Neumora, Neurocrine Biosciences, Noven, Otsuka, Sensorium Therapeutics, Sirtsei Pharmaceuticals, Supernus, Teva, Thynk, Transneural Therapeutics, Tris Pharma, Vanda Pharmaceuticals, and Vistagen. He is on a speaker's bureau for AbbVie, Alfasigma, Alkermes, Axsome, Aytu Biopharma, Bristol Myers Squibb, Collegium Pharmaceutical, Intra-Cellular Therapies, J&J Innovative Medicine, Knight Therapeutics, Kye Pharmaceuticals, Lundbeck, Luye Pharma, Neurocrine Biosciences, Noven, Otsuka, Supernus, Teva, Tris Pharma, and Vanda Pharmaceuticals; a Data Safety Monitoring Board for Compass Pathways and holds stock options/equity in EMA Wellness, Evolution Research Group, 4M Therapeutics, Sensorium Therapeutics, and Transneural Therapeutics. **Rakesh Jain** is a consultant, researcher, paid speaker and on an advisory board for Lilly, Lundbeck, Otsuka, Pfizer, Shire, and Takeda; a consultant, paid speaker and on an advisory board for Alkermes, Corium, Eisai, Johnson & Johnson, Merck, Neos Therapeutics, Pamlab, and Sunovion; a consultant, researcher, and is a paid speaker for AbbVie; a consultant and on an advisory board for Adamas, Neurocrine Biosciences, Sage Therapeutics, Supernus, and Teva; a consultant and paid speaker for Almatica, Axsome, Sumitomo, and Viatris; a consultant for Acadia, Alfasigma, Biogen, Boehringer Ingleheim, Cingulate Therapeutics, Evidera, Impel, Osmotica, and Transcend Therapeutics; on an advisory board for Usono, and a paid speaker for Intra-Cellular Therapies, Ironshore Pharmaceuticals, and Tris Pharmaceuticals. **Katharine Henry, Dana Drzayich Jankus, Yordan Godinov, Filip Delaei, Onur Tankaya, Katalin Pungor,** and **Dong-Jing Fu** are employees of Johnson & Johnson and may hold stock/stock options in Johnson & Johnson.

Novel Pathways in Depression

Scan the QR code

The QR code is intended to provide scientific information for individual reference, and the information should not be altered or reproduced in any way.

Presented at: Psych Congress 2026, September 15–19, 2026, New Orleans, LA.

REFERENCES:
1. Wong S, Le GH, Phan L, et al. *J Affect Disord.* 2024;356:684-698. **2.** Cutler AJ, Clayton AH, Krystal AD, et al. *Psychiatry Res.* 2026;364:117225. **3.** Spravato® (esketamine nasal spray) prescribing information. Johnson & Johnson; 2026. **4.** Pepe M, Bartolucci G, Marcelli I, et al. *Brain Sci.* 2023;13:1494. **5.** Fu DJ, Bossie C, Tankaya O, et al. Poster presented at 64th Annual ACNP. Nassau, Bahamas; January 12–15, 2026. **6.** Wu C, Mu Q, Gao W, et al. *Transl Psychiatry.* 2025;15:90.