96

Efficacy of Esketamine Nasal Spray Monotherapy for Treatment-Resistant Depression: Post Hoc Analysis by Baseline Disease Severity and Prior Treatment

Mai Himedan[1], Oliver Lopena[1], Lisa Lim[1], Alexis Davis[1], Ifeanyi Valentine Uche[1], Dong-Jing Fu[1], Ibrahim Turkoz[1], Michael Asbach[2]

[1]Johnson & Johnson, Titusville, NJ; [2]DENT Neurologic Institute, Buffalo, NY

Introduction

- Patients with major depressive disorder who have not benefitted from 2 or more prior antidepressants (AD) despite adequate dose and duration in the current depressive episode are considered to have treatment-resistant depression (TRD). This population experiences disproportionately higher morbidity and mortality associated with depression[1]
- Esketamine nasal spray (ESK) is approved by the US Food and Drug Administration for the treatment of TRD in adults, as monotherapy or in conjunction with an oral AD, and for the treatment of depressive symptoms in adults with major depressive disorder with acute suicidal ideation or behavior, in conjunction with an oral AD[2]
- A randomized, double-blind, placebo-controlled, multicenter study (NCT04599855) demonstrated that ESK monotherapy resulted in significant and clinically meaningful improvements in depressive symptoms compared with placebo at day 28, and as early as 24 hours, in adults with TRD, and that safety and tolerability were consistent with the established safety profile of ESK[3]

Objective

- This post hoc analysis evaluated the efficacy and safety of ESK monotherapy in adults with TRD by baseline disease severity and number of prior unsuccessful AD treatments

Methods

Study design and assessments

- This was a phase 4, double-blind, randomized, placebo-controlled study conducted at multiple sites in the US (**Figure 1**)
 - During the screening phase, patients were required to complete a minimum of 2 weeks without any AD medication just prior to randomization
 - In the double-blind treatment phase, eligible patients who met a predefined severity criterion were randomized 2:1:1 to receive placebo, ESK 56 mg, or ESK 84 mg twice a week for 4 weeks
- Efficacy was assessed by Montgomery-Åsberg Depression Rating Scale (MADRS) total score (range 0-60, with higher scores indicating more severe depression) on days 2 (24 hours post dose), 8, 15, 22, and 28
- Safety was assessed by monitoring treatment-emergent adverse events (TEAEs) throughout the study

Analyses

- Analyses were performed for the following subgroups:
 - Baseline MADRS total score ≤ or > than the overall median baseline MADRS total score
 - Number of prior unsuccessful AD treatments (2 or ≥3)
- Changes in MADRS total score from baseline to day 28 were analyzed using a mixed model for repeated measures (MMRM). The model included fixed effects for treatment (ESK 56 mg, ESK 84 mg, placebo), visit (categorical), treatment-by-visit interaction, and baseline MADRS total score as covariate in addition to additional factors
 - Least squares (LS) mean changes from baseline and LS mean differences versus placebo with two-sided 95% confidence intervals (CIs) are shown for each ESK dose versus placebo
 - A 6-point improvement in MADRS total score was considered a clinically meaningful change within a patient[4]
- The proportions of patients who achieved response (≥50% reduction from baseline in MADRS total score and remission (MADRS total score ≤12) are summarized at day 28

Figure 1: Study design

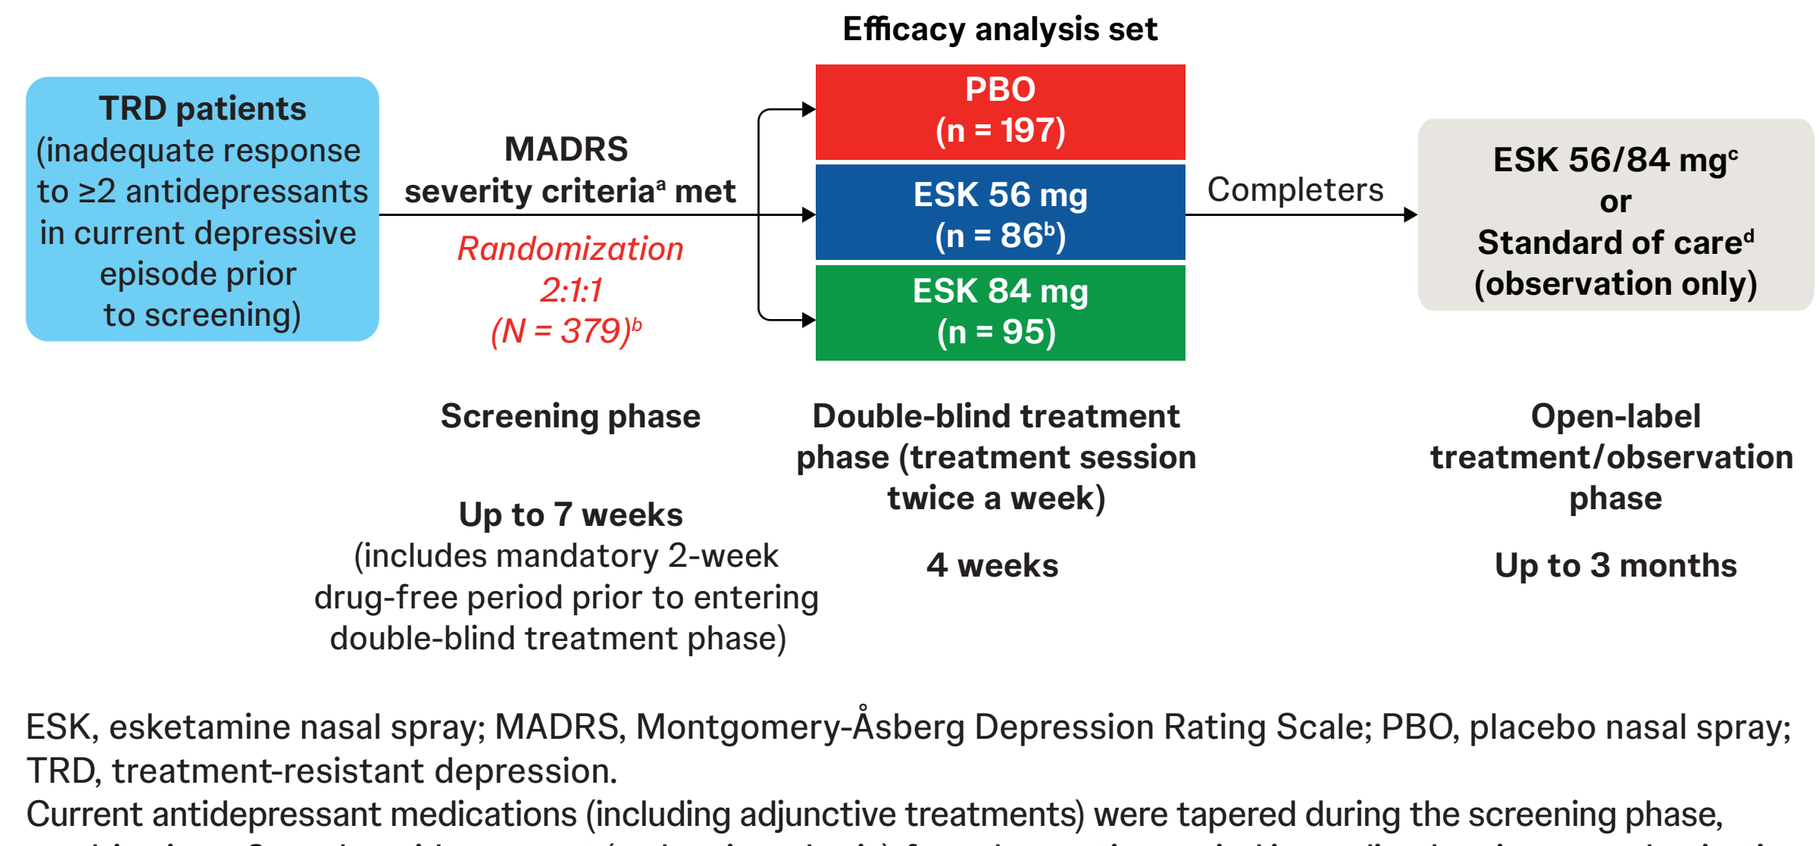
Efficacy analysis set
TRD patients (inadequate response to ≥2 antidepressants in current depressive episode prior to screening)
MADRS severity criteria[a] met
Randomization 2:1:1 (N = 379)[b]
PBO (n = 197)
ESK 56 mg (n = 86[b])
ESK 84 mg (n = 95)
Completers
ESK 56/84 mg[c] or Standard of care[d] (observation only)
Screening phase
Up to 7 weeks (includes mandatory 2-week drug-free period prior to entering double-blind treatment phase)
Double-blind treatment phase (treatment session twice a week)
4 weeks
Open-label treatment/observation phase
Up to 3 months

ESK, esketamine nasal spray; MADRS, Montgomery-Åsberg Depression Rating Scale; PBO, placebo nasal spray; TRD, treatment-resistant depression.
Current antidepressant medications (including adjunctive treatments) were tapered during the screening phase, resulting in a ≥2-week antidepressant (and antipsychotic)-free observation period immediately prior to randomization.
[a]MADRS total score ≥28 at screening week 1, week 2, and day 1 (pre-randomization) and ≤25% improvement in the MADRS total score from screening week 1 to day 1 (pre-randomization); referred to as "nonresponse criteria" in the protocol.
[b]One of 87 patients randomly assigned to ESK 56 mg did not receive a dose of study drug and was not included in the efficacy analysis set.
[c]With or without standard of care.
[d]Only 1 patient received standard of care without ESK.

Results

Demographics and baseline characteristics

- A total of 378 adults with TRD were randomly assigned to receive ESK 56 mg, ESK 84 mg, or placebo in the double-blind treatment phase; the baseline median MADRS total score for overall patients was 37.0
- More than half of patients (52.6%) had baseline MADRS total score ≤ the overall baseline median MADRS total score of 37 (**Table 1**), and 59.3% of patients had 2 prior unsuccessful AD treatments (**Table 2**)
- Demographics were balanced across treatment groups and subgroups; most patients were female (range 53.5%-65.5%) and White (range 81.1%-93.9%), and mean patient age ranged from 43.1 to 49.8 years

Efficacy by baseline MADRS total score

- Regardless of baseline MADRS total score, treatment with ESK was associated with clinically meaningful improvements in MADRS total score compared with placebo from baseline to day 28 (**Figure 2**)
- Patients treated with ESK 56 mg and ESK 84 mg achieved higher rates of response (≥50% improvement from baseline in MADRS total score) and remission (MADRS total score ≤12) compared with placebo at every time point measured, regardless of baseline disease severity (day 28 shown in **Figure 3**)

Efficacy by number of prior antidepressant treatments

- ESK of either dose was associated with clinically meaningful improvements from baseline to day 28 in MADRS total score compared with placebo (**Figure 4**)
- Patients treated with either dose of ESK had higher rates of response (≥50% improvement from baseline in MADRS total score) and remission (MADRS total score ≤12) compared with placebo at all time points, regardless of number of prior AD treatments (day 28 shown in **Figure 5**)

Safety

- Similar to results from the overall study population, the most common reported TEAEs were nausea, dissociation, dizziness, and headache (**Table 3**)[3]
- Commonly reported TEAEs occurred more frequently in the ESK groups than with placebo
- The percentages of patients who withdrew from the study due to TEAEs were <6% for all treatment groups, regardless of baseline MADRS total score or number of prior AD treatments

Table 1: Patient demographics and baseline clinical characteristics by baseline MADRS total score

	Baseline MADRS total score ≤37 n = 199			Baseline MADRS total score >37 n = 179		
	ESK 56 mg n = 43	ESK 84 mg n = 52	PBO n = 104	ESK 56 mg n = 43	ESK 84 mg n = 43	PBO n = 93
Mean age (SD), years	49.8 (14.28)	44.5 (16.33)	45.3 (14.63)	43.1 (13.41)	45.1 (12.49)	45.2 (12.81)
Female, n (%)	23 (53.5)	34 (65.4)	62 (59.6)	28 (65.1)	27 (62.8)	57 (61.3)
Race, n (%)						
Asian	0	1 (1.9)	3 (2.9)	2 (4.7)	3 (7.0)	2 (2.2)
Black or African American	2 (4.7)	6 (11.5)	4 (3.8)	2 (4.7)	2 (4.7)	9 (9.7)
White	39 (90.7)	43 (82.7)	94 (90.4)	37 (86.0)	38 (88.4)	77 (82.8)
Other[a]	2 (4.7)	2 (3.8)	3 (2.9)	2 (4.7)	0	5 (5.5)
Ethnicity, n (%)						
Hispanic or Latino	4 (9.3)	4 (7.7)	13 (12.5)	4 (9.3)	2 (4.7)	4 (4.3)
Mean duration of current episode (range), weeks	400.3 (22-2340)	452.2 (10-2236)	282.6 (10-1716)	439.3 (12-2555)	351.1 (19-1456)	296.2 (12-1872)
Number of episodes since diagnosis, n (%)						
≥3	30 (69.8)	28 (53.8)	60 (57.7)	24 (55.8)	27 (62.8)	67 (72.0)
History of suicidal ideation in past 6 months, n (%)	13 (30.2)	29 (55.8)	50 (48.1)	25 (58.1)	23 (53.5)	55 (59.1)
Mean baseline CGI-S score (SD)	4.8 (0.51)	4.7 (0.52)	4.7 (0.53)	5.2 (0.63)	5.2 (0.72)	5.2 (0.61)
Mean baseline MADRS total score (SD)	33.2 (2.78)	33.2 (2.16)	33.7 (2.76)	41.7 (3.24)	40.7 (2.72)	41.8 (2.83)

CGI-S, Clinical Global Impression-Severity; ESK, esketamine nasal spray; MADRS, Montgomery-Åsberg Depression Rating Scale; PBO, placebo nasal spray.
[a]Includes American Indian, Alaska Native, Native Hawaiian or other Pacific Islander, Multiple, Not Reported, and Unknown.

Table 2: Patient demographics and baseline clinical characteristics by number of prior AD treatments

	Number of prior AD treatments =2 n = 224			Number of prior AD treatments ≥3 n = 154		
	ESK 56 mg n = 49	ESK 84 mg n = 58	PBO n = 117	ESK 56 mg n = 37	ESK 84 mg n = 37	PBO n = 80
Mean age (SD), years	45.9 (13.47)	43.6 (13.34)	46.6 (13.21)	47.2 (15.23)	46.7 (16.50)	43.3 (14.39)
Female, n (%)	30 (61.2)	38 (65.5)	73 (62.4)	21 (56.8)	23 (62.2)	46 (57.5)
Race, n (%)						
Asian	2 (4.1)	4 (6.9)	3 (2.6)	0	0	2 (2.5)
Black or African American	0	6 (10.3)	7 (6.0)	4 (10.8)	2 (5.4)	6 (7.5)
White	46 (93.9)	48 (82.8)	103 (88.0)	30 (81.1)	33 (89.2)	68 (85.0)
Other[a]	1 (2.0)	0	4 (3.5)	3 (8.1)	2 (5.4)	4 (5.0)
Ethnicity, n (%)						
Hispanic or Latino	5 (10.2)	3 (5.2)	12 (10.3)	3 (8.1)	3 (8.1)	5 (6.3)
Mean duration of current episode (range), weeks	305.3 (22-2340)	232.0 (10-1196)	248.3 (10-1872)	571.5 (12-2555)	679.8 (20-2236)	348.5 (12-1414)
Number of episodes since diagnosis, n (%)						
≥3	33 (67.3)	41 (70.7)	82 (70.1)	21 (56.8)	14 (37.8)	45 (56.3)
History of suicidal ideation in past 6 months, n (%)	22 (44.9)	31 (53.4)	52 (44.4)	16 (43.2)	21 (56.8)	53 (66.3)
Mean baseline CGI-S score (SD)	4.9 (0.56)	5.0 (0.57)	4.9 (0.59)	5.1 (0.66)	4.9 (0.77)	4.9 (0.63)
Mean baseline MADRS total score (SD)	37.2 (5.23)	36.6 (4.47)	37.5 (5.08)	37.8 (5.29)	36.5 (4.56)	37.6 (4.66)

AD, antidepressant; CGI-S, Clinical Global Impression-Severity; ESK, esketamine nasal spray; MADRS, Montgomery-Åsberg Depression Rating Scale; PBO, placebo nasal spray.
[a]Includes American Indian, Alaska Native, Native Hawaiian or other Pacific Islander, Multiple, Not Reported, and Unknown.

Figure 2: Mean change in MADRS total score from baseline to day 28 by baseline MADRS total score

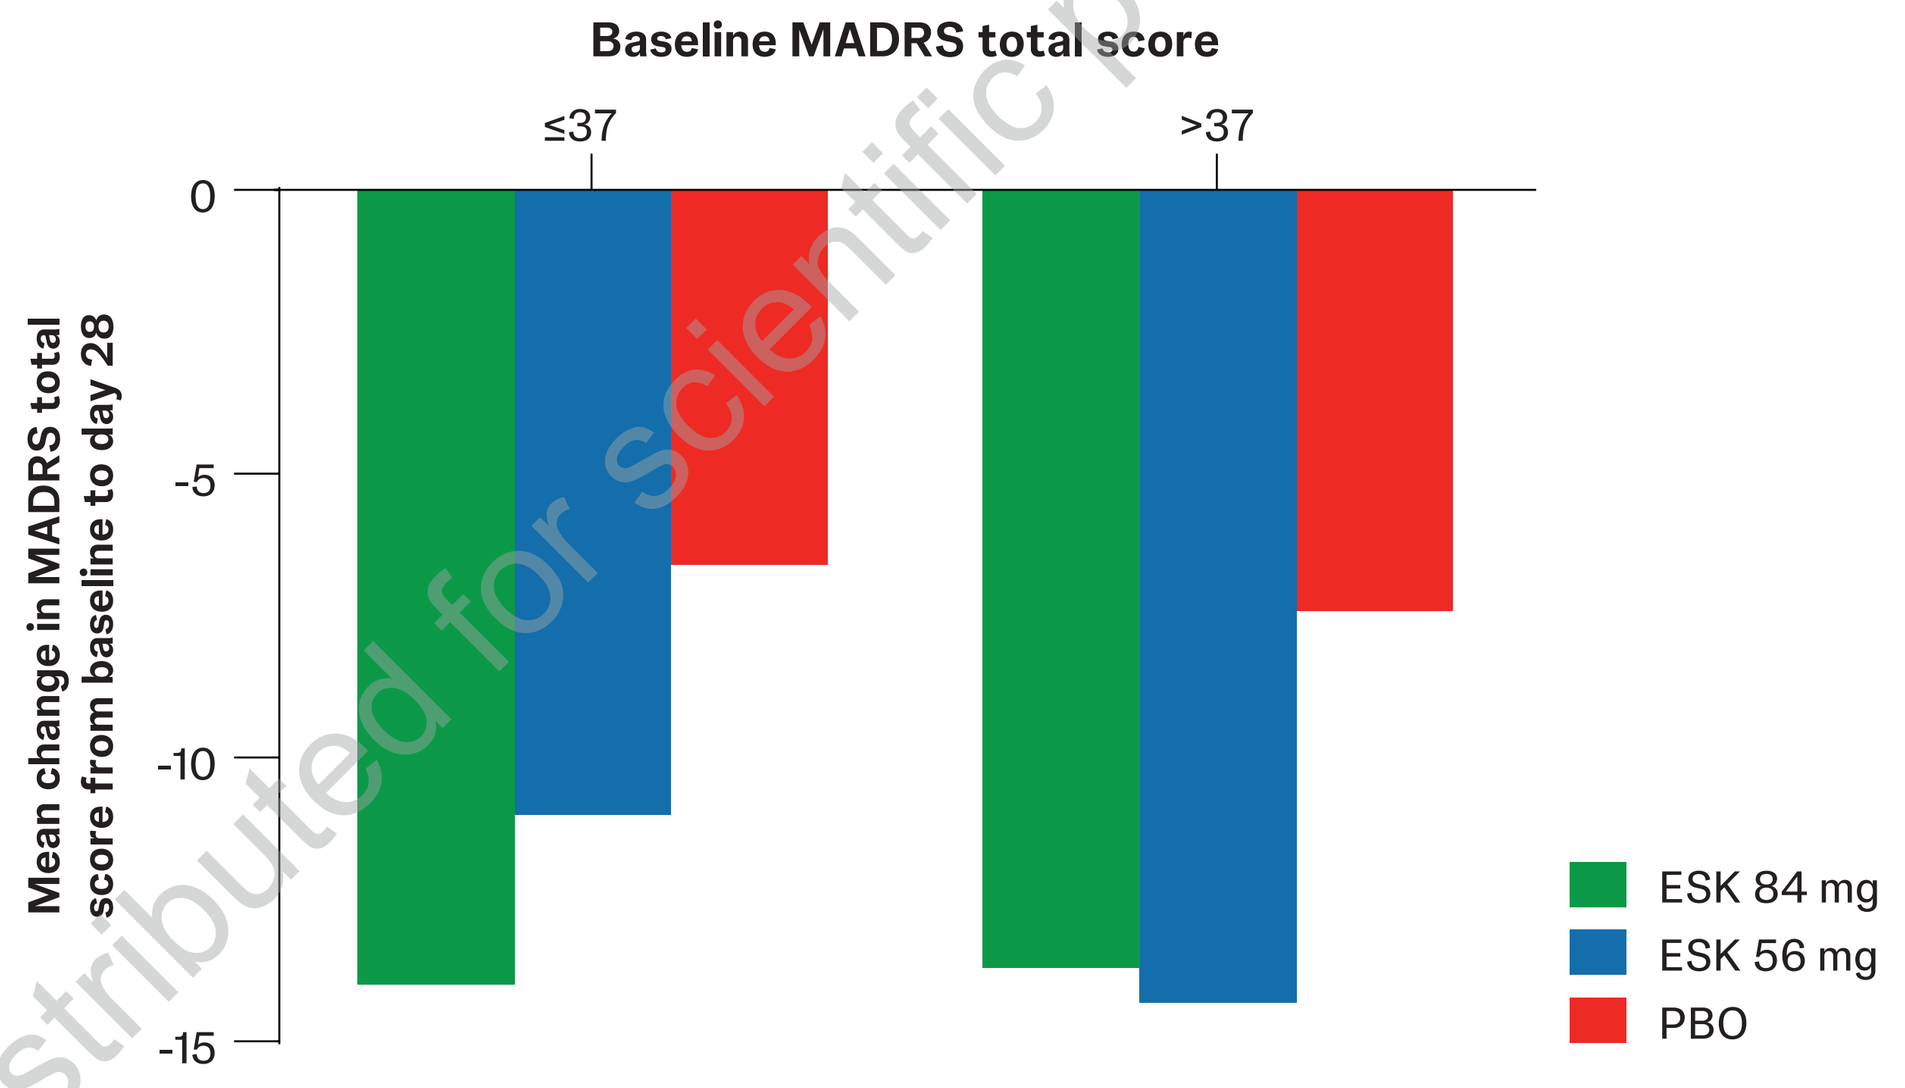
Baseline MADRS total score
≤37
>37
0
-5
-10
-15
Mean change in MADRS total score from baseline to day 28
ESK 84 mg
ESK 56 mg
PBO

LS mean difference (95% CI) from PBO

	≤37	>37
ESK 56 mg	-3.7 (-7.72, 0.28)	-6.4 (-10.39, -2.42)
ESK 84 mg	-7.9 (-11.59, -4.13)	-5.3 (-9.41, -1.27)

ESK, esketamine nasal spray; LS, least squares; MADRS, Montgomery-Åsberg Depression Rating Scale; PBO, placebo nasal spray.

Figure 3: Proportion of patients achieving (A) response or (B) remission per MADRS total score on day 28 by baseline MADRS total score

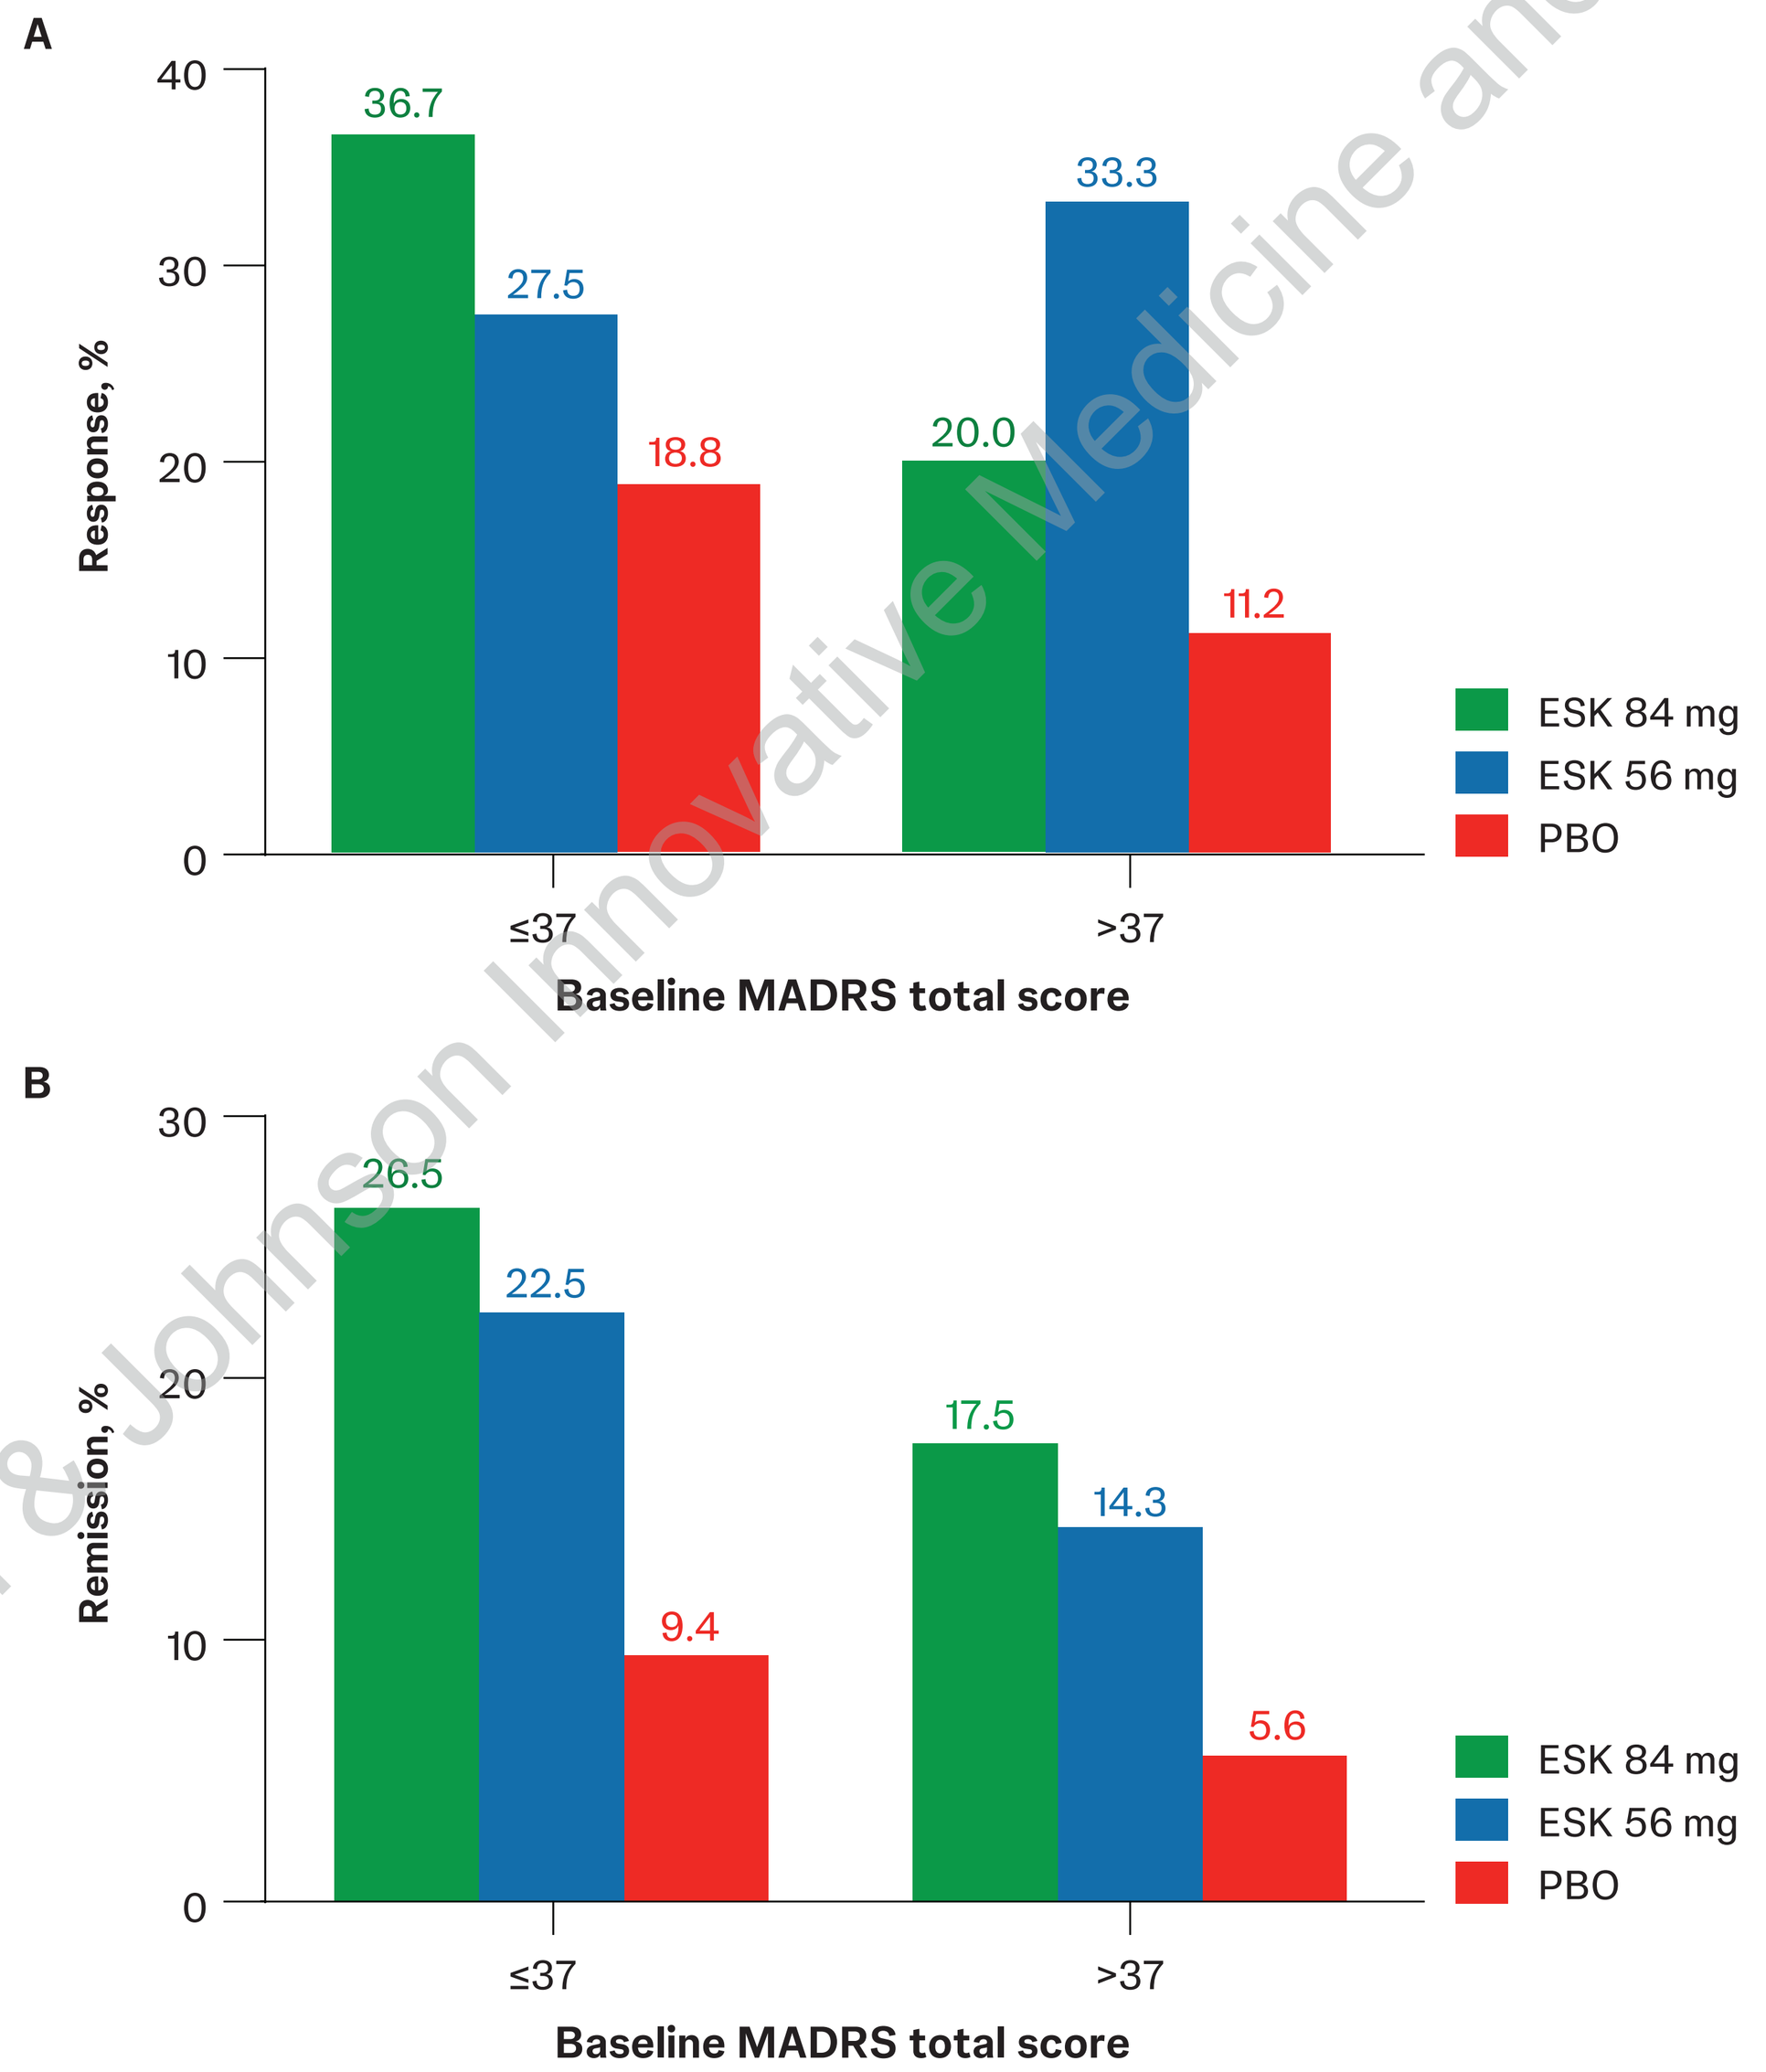
A
Response, %
40
30
20
10
0
36.7
27.5
18.8
20.0
33.3
11.2
≤37
>37
Baseline MADRS total score
ESK 84 mg
ESK 56 mg
PBO
B
Remission, %
30
20
10
0
26.5
22.5
9.4
17.5
14.3
5.6
≤37
>37
Baseline MADRS total score
ESK 84 mg
ESK 56 mg
PBO

ESK, esketamine nasal spray; MADRS, Montgomery-Åsberg Depression Rating Scale; PBO, placebo nasal spray.
Response is defined as a ≥50% improvement in MADRS total score. Remission is defined as a MADRS total score ≤12.

Figure 4: Mean change in MADRS total score from baseline to day 28 by number of prior AD treatments

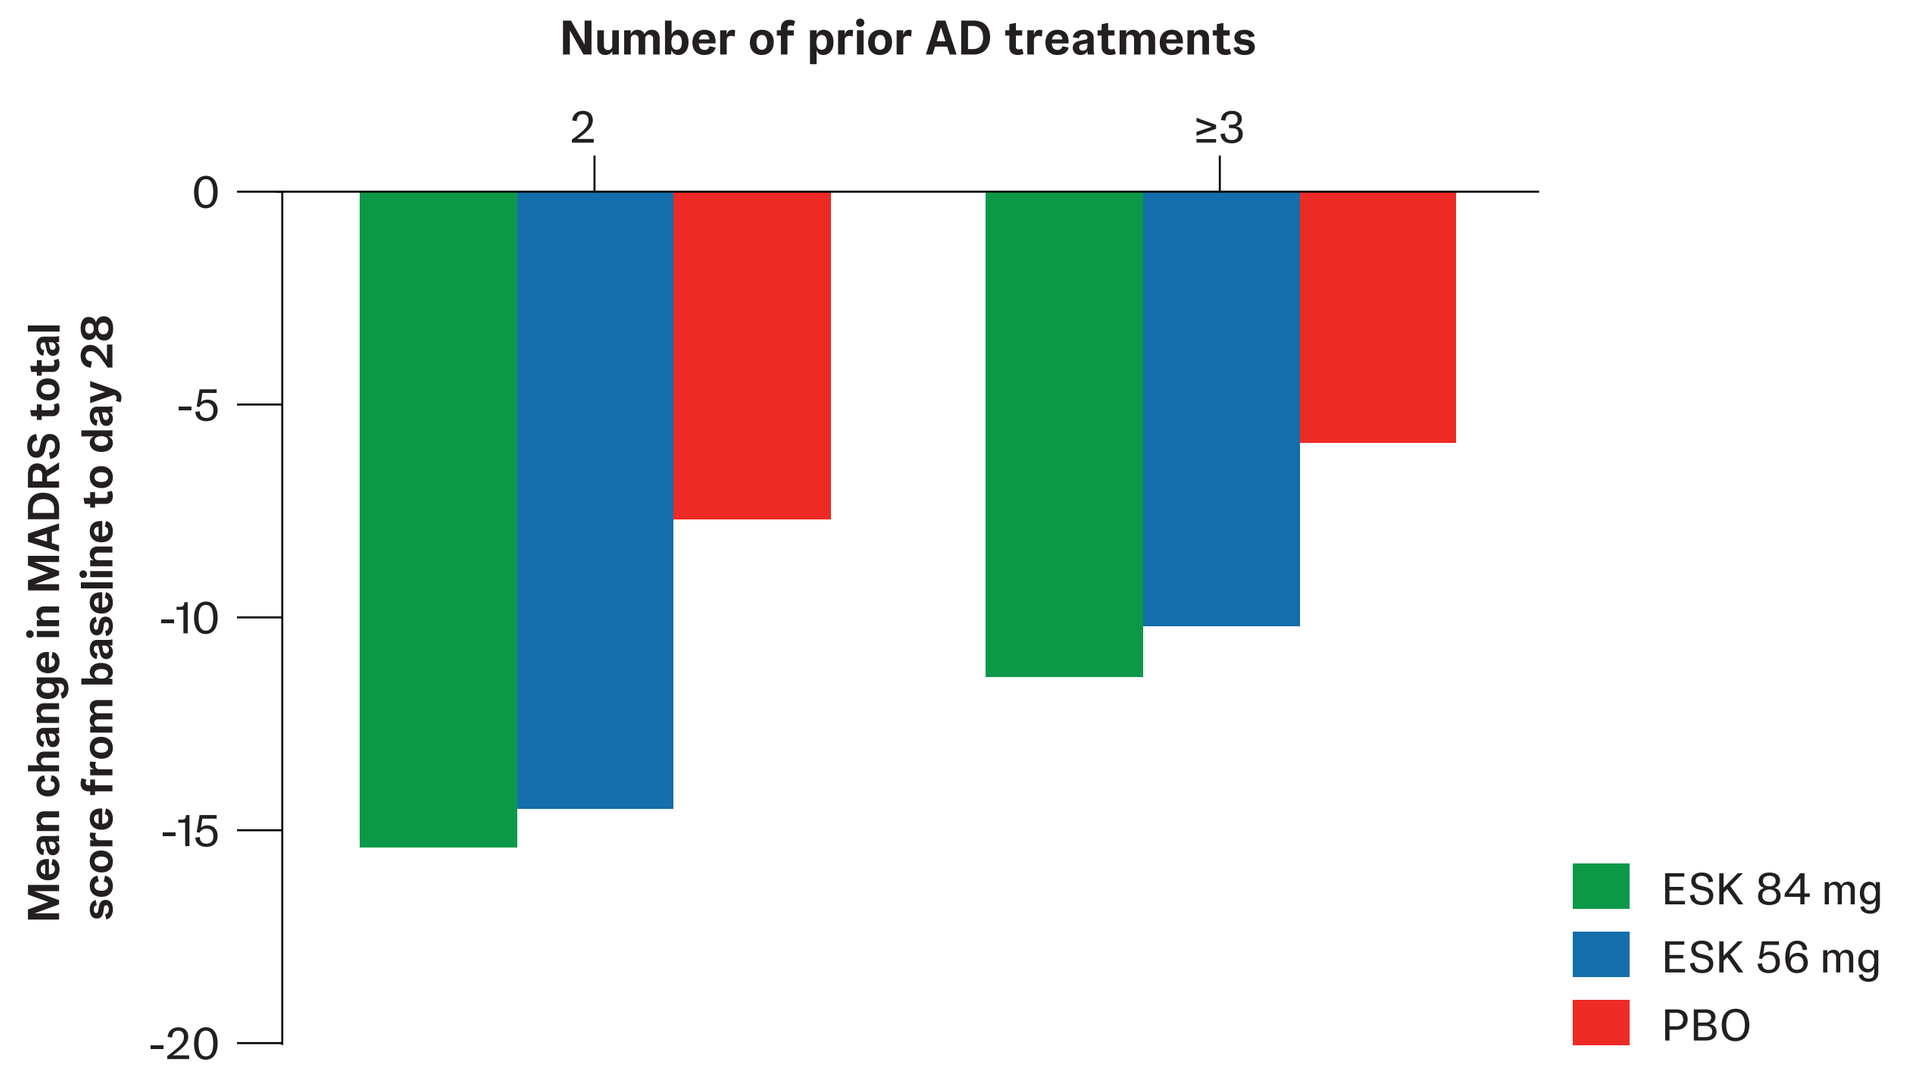
Number of prior AD treatments
2
≥3
0
-5
-10
-15
-20
Mean change in MADRS total score from baseline to day 28
ESK 84 mg
ESK 56 mg
PBO

LS mean difference (95% CI) from PBO

	2	≥3
ESK 56 mg	-5.3 (-9.03, -1.65)	-5.0 (-9.39, -0.60)
ESK 84 mg	-7.4 (-10.93, -3.90)	-5.7 (-10.12, -1.29)

AD, antidepressant; ESK, esketamine nasal spray; LS, least squares; MADRS, Montgomery-Åsberg Depression Rating Scale; PBO, placebo nasal spray.

Figure 5: Proportion of patients achieving (A) response or (B) remission per MADRS total score on day 28 by number of prior unsuccessful AD treatments

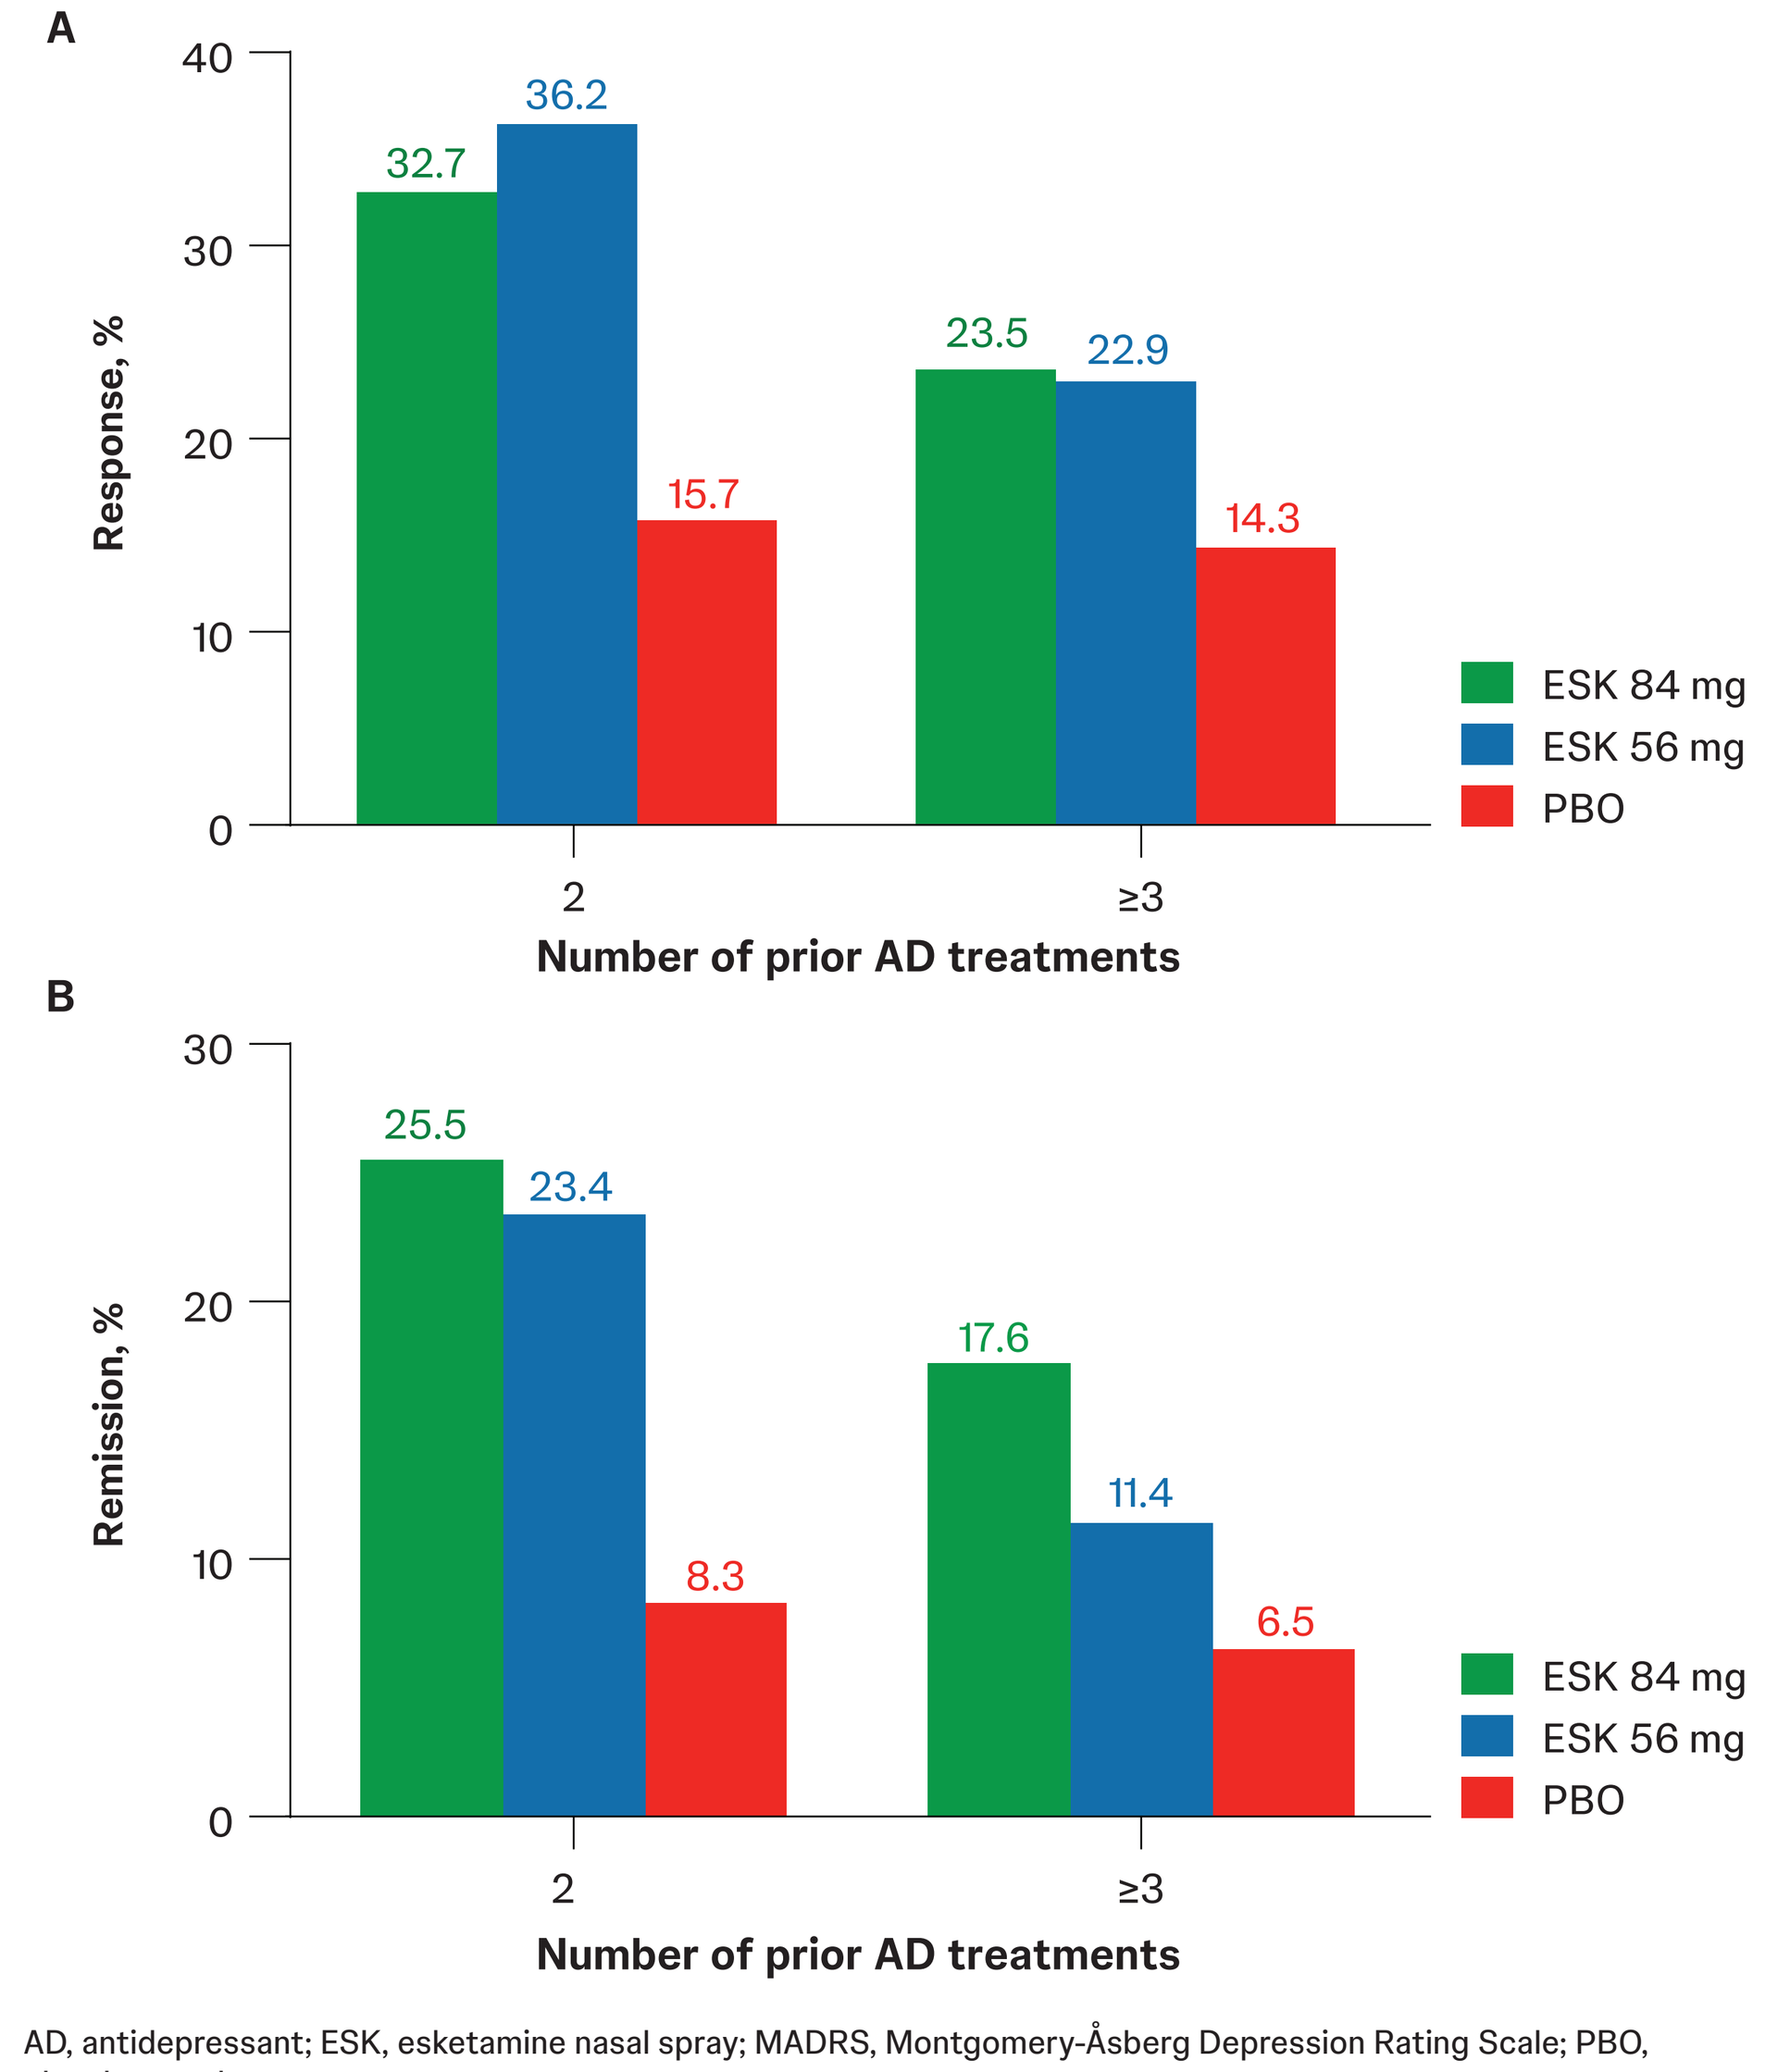
A
Response, %
40
30
20
10
0
32.7
36.2
15.7
23.5
22.9
14.3
2
≥3
Number of prior AD treatments
ESK 84 mg
ESK 56 mg
PBO
B
Remission, %
30
20
10
0
25.5
23.4
8.3
17.6
11.4
6.5
2
≥3
Number of prior AD treatments
ESK 84 mg
ESK 56 mg
PBO

AD, antidepressant; ESK, esketamine nasal spray; MADRS, Montgomery-Åsberg Depression Rating Scale; PBO, placebo nasal spray.
Response is defined as a ≥50% improvement in MADRS total score. Remission is defined as a MADRS total score ≤12.

Table 3: Most common TEAEs (≥10%)

	Baseline MADRS total score ≤37			Baseline MADRS total score >37		
TEAEs, n (%)	ESK 56 mg n = 43	ESK 84 mg n = 52	PBO n = 104	ESK 56 mg n = 43	ESK 84 mg n = 43	PBO n = 93
Nausea	12 (27.9)	14 (26.9)	13 (12.5)	10 (23.3)	10 (23.3)	5 (5.4)
Dissociation	8 (18.6)	15 (28.8)	2 (1.9)	11 (25.6)	9 (20.9)	5 (5.4)
Dizziness	11 (25.6)	9 (17.3)	8 (7.7)	8 (18.6)	13 (30.2)	7 (7.5)
Headache	6 (14.0)	9 (17.3)	9 (8.7)	7 (16.3)	10 (23.3)	8 (8.6)
Vomiting	2 (4.7)	4 (7.7)	0	3 (7.0)	5 (11.6)	1 (1.1)
	Number of prior AD treatments =2			**Number of prior AD treatments ≥3**		
TEAEs, n (%)	ESK 56 mg n = 49	ESK 84 mg n = 58	PBO n = 117	ESK 56 mg n = 37	ESK 84 mg n = 37	PBO n = 80
Nausea	10 (20.4)	14 (24.1)	10 (8.5)	12 (32.4)	10 (27.0)	8 (10.0)
Dissociation	10 (20.4)	13 (22.4)	5 (4.3)	9 (24.3)	11 (29.7)	2 (2.5)
Dizziness	11 (22.4)	8 (13.8)	9 (7.7)	8 (21.6)	14 (37.8)	6 (7.5)
Headache	5 (10.2)	9 (15.5)	10 (8.5)	8 (21.6)	10 (27.0)	7 (8.8)
Vomiting	2 (4.1)	5 (8.6)	1 (0.9)	3 (8.1)	4 (10.8)	0
Feeling drunk	1 (2.0)	2 (3.4)	1 (0.9)	6 (16.2)	4 (10.8)	1 (1.3)
Fatigue	3 (6.1)	3 (5.2)	6 (5.1)	4 (10.8)	2 (5.4)	5 (6.3)
Euphoric mood	0	0	2 (1.7)	0	4 (10.8)	0

AD, antidepressant; ESK, esketamine nasal spray; MADRS, Montgomery-Åsberg Depression Rating Scale; PBO, placebo nasal spray; TEAE, treatment-emergent adverse event.

Key Takeaway

Esketamine nasal spray monotherapy led to clinically meaningful improvements in depressive symptoms in patients with treatment-resistant depression, regardless of baseline disease severity or number of prior unsuccessful antidepressant treatments

Limitations

The generalizability of these findings may be limited by the exclusion of patients with significant psychiatric or medical comorbidities, or substance dependence

These subgroup analyses were not prespecified and small sample sizes limit the ability to perform statistical comparison

Conclusions

Patients with treatment-resistant depression who received esketamine nasal spray monotherapy had clinically meaningful improvements in MADRS total score and higher rates of response and remission compared with placebo nasal spray, regardless of baseline disease severity or number of prior unsuccessful antidepressant treatments

Reported treatment-emergent adverse events were consistent with the established safety and tolerability profile for esketamine nasal spray

Acknowledgements
The authors thank Emily Spaulding, PhD (ApotheCom, Yardley, PA), for editorial and writing assistance, which was funded by Johnson & Johnson.

Disclosures
MH, OL, LL, AD, IVU, D-JF, and IT are employees of Johnson & Johnson and may hold stock or restricted stock units in the company. MA has served as a speaker and/or consultant for Bristol Myers Squibb, Intracellular Therapies Inc., Biogen, Axsome Therapeutics, Janssen Pharmaceuticals, Otsuka Pharmaceuticals, Abbvie, Neurocrine Biosciences, Avanir Pharmaceuticals, and Karuna Therapeutics.

Previously presented at the Neuroscience Education Institute (NEI) Spring Congress; May 1-3, 2026; Kissimmee, Florida, USA

Novel Pathways in Depression

Scan the QR code

The QR code is intended to provide scientific information for individual reference, and the information should not be altered or reproduced in any way.

This study was funded by Johnson & Johnson.

Presented at Psych Congress; September 15-19, 2026; New Orleans, Louisiana, USA

References
1. McIntyre RS et al. *World Psychiatry*. 2023;22(3):394-412. 2. SPRAVATO® (esketamine) nasal spray, CIII. Prescribing information. Janssen Pharmaceuticals, Inc.; 2025:16. Accessed August 12, 2026. https://www.janssenlabels.com/package-insert/product-monograph/prescribing-information/SPRAVATO-pi.pdf 3. Janik A et al. *JAMA Psychiatry*. 2025;82(9):877-887. 4. Turkoz I et al. *Acta Psychiatr Scand*. 2021;143(3):253-263.

This material is distributed for scientific purposes by Johnson & Johnson Innovative Medicine and is not for promotional use.