15

Association of Schizophrenia-Related Hospitalization Before Long-Acting Injectable Antipsychotic Initiation and Outcomes Among Young Dual Eligibles

Pengxiang Li[1], Zhi Geng[1], Carmela Benson[2], Jalpa A. Doshi[1,3]

[1]Division of General Internal Medicine, University of Pennsylvania, Philadelphia, PA; [2]Johnson & Johnson, Titusville, NJ; [3]Leonard Davis Institute of Health Economics, Philadelphia, PA

This material is distributed for scientific purposes by Johnson & Johnson Innovative Medicine and is not for promotional use.

Background

- Schizophrenia (SCZ) is a chronic, severe psychiatric disorder associated with substantial clinical, humanistic, and economic burden[1]
- Suboptimal adherence to oral antipsychotics (APs) is common and is a major contributor to relapses, emergency department visits, and SCZ-related hospitalizations[2–4]
- Real-world evidence has demonstrated that long-acting injectable antipsychotics (LAIs) are associated with improved medication adherence and a reduced risk of SCZ-related hospitalizations compared with oral APs[5,6]
- However, LAI initiation is often delayed in young people with SCZ[7]

Objective

- The CRITICAL PERIOD study intended to examine the SCZ-related hospitalization history prior to initiating an LAI and its association with adherence, discontinuation, relapse, and healthcare resource use in young dual-eligible Medicare beneficiaries with SCZ in the United States

Methods

Data source

- Data were taken from 2009 to 2021 national Medicare and Medicaid fee-for-service (FFS) claims available from the Centers for Medicare and Medicaid Services

Sample selection

- All beneficiaries having ≥1 claim with International Classification of Diseases (ICD)-9 code or ICD-10 code of SCZ in any position from the Medicaid or Medicare FFS claims, with the first observed claims occurring between 7/1/2009 and 12/31/2021
- Dually eligible for Medicare and Medicaid when they were first eligible for Medicare
- ≥1 antipsychotic fill between July 1, 2009, and December 31, 2021, with the first AP fill occurring on or after the date of the first observed SCZ diagnosis date in Medicare or Medicaid claims
- Continuous full Medicaid (FFS or health maintenance organization [HMO]) coverage or Medicare FFS Part D coverage from first SCZ diagnosis date till first AP fill date (i.e. index date)
- First eligible for Medicare due to disability (excluding patients aging into Medicare since they likely had private insurance prior to turning 65 years old)
- Use of an LAI between July 1, 2009, and December 31, 2021; the first date of LAI use was the index date
- Time between the first observed SCZ diagnosis date and first LAI use date (index date) was 1 month or more
- Continuous coverage of at least 12 months in the post-index period (to allow a 12-month fixed follow-up period to measure outcomes)
- Age ≤25 years when first diagnosed with schizophrenia

Independent variable

- Patients were stratified by the number of SCZ-related hospitalizations in the pre-index period (0, 1, 2-5, or ≥6 hospitalizations)

Outcomes

- **Adherence:** Defined as proportion of days covered (PDC) ≥0.80 for any LAI or any AP during the 12-month post-index period
- **Discontinuation:** Defined as a ≥60-day continuous gap in days' supply for any LAI or any AP during the 12-month post-index period
- **Evidence of relapse:** Identified based on claims with a schizophrenia diagnosis in first or second diagnosis position on inpatient or emergency room visit claims during the 12-month post-index period
- **Any hospitalizations** during the 12-month post-index period
 - All-cause hospitalizations
 - Mental health-related hospitalizations
 - SCZ-related hospitalizations

Analysis

- Descriptive analyses were conducted on patient characteristics and outcomes in the overall sample and by independent variable
- Multivariable logistic regression models were used to assess the association between pre-index hospitalization category and adherence, discontinuation, relapse, and any hospitalizations during the 12-month follow up period, while controlling demographic and clinical characteristics

Results

- The final sample consisted of 3,447 patients initiating an LAI (**Table 1**)
- Patients had similar mean age of 22 years on SCZ diagnosis date across groups; they were primarily male (78%) and non-White (62%, **Table 2**)
 - Over half (51%) of patients had 0 SCZ-related hospitalizations in the pre-index period; the remaining patients had 1 (22%), 2 to 5 (22%), and ≥6 (5%) SCZ-related hospitalizations, respectively
 - The median time from first observed SCZ diagnosis to first observed LAI initiation was 331 days (162 for 0, 373 for 1, 719 for 2 to 5, and 1251 for ≥6 SCZ-related hospitalizations)
- The most frequently used LAIs were paliperidone (46%), haloperidol (17%), and aripiprazole (16%, **Table 3**)
- In descriptive analyses, increasing numbers of prior hospitalizations were associated with lower adherence to any LAI (35% among patients with 0 prior hospitalizations vs. 22% among those with ≥6 prior hospitalizations), higher discontinuation of any LAI (53% vs. 70%), higher relapse rates (49% vs. 73%), and higher rates of all-cause (28% vs. 71%), mental health-related (26% vs. 71%), and schizophrenia-related hospitalizations (22% vs. 68%) during follow-up (**Table 4**)
- Logistic regressions confirmed these results (**Table 5**)
 - For instance, patients with ≥6 vs 0 pre-index hospitalizations had lower odds of LAI adherence (odds ratio [OR] 0.49; *P* < 0.01) and higher odds of relapse (OR 2.73; *P* < 0.01) and hospitalization (all-cause OR 3.92; MH-related OR 4.20; SCZ-related OR 4.93; all *P* < 0.01)

Table 1: Sample attrition

Criteria	N
All beneficiaries having ≥1 claim with ICD-9 code or ICD-10 code of SCZ in any position from the Medicaid or Medicare FFS claims, with the first observed claims occurring between July 1, 2009, and December 31, 2021	1,338,215
Dually eligible for Medicare and Medicaid when they were first eligible for Medicare	340,524
≥1 antipsychotic fill between July 1, 2009, and December 31, 2021, with the first AP fill date occurring on or after the date of the first observed SCZ diagnosis date in Medicare or Medicaid claims	86,110
Continuous full Medicaid (FFS or HMO) coverage or Medicare FFS Part D coverage from first SCZ diagnosis date till first AP fill date (i.e. index date)	40,412
First eligible for Medicare due to disability (excluding patients aging into Medicare since they likely had private insurance prior to turning 65 years old)	33,102
Use of an LAI between July 1, 2009, and December 31, 2021; the first date of LAI use was the index date	11,735
Time between the first observed SCZ diagnosis date and first LAI use date (index date) was 1 month or more	10,808
Continuous coverage of at least 12 months in the post-index period (to allow a 12-month fixed follow-up period to measure outcomes)	9,472
Age ≤25 years when first diagnosed with schizophrenia	3,447

AP, antipsychotic; FFS, fee for service; HMO, health maintenance organization; ICD, International Classification of Diseases; LAI, long-acting injectable; SCZ, schizophrenia.

Table 2: Patient characteristics

		Number of schizophrenia-related hospitalizations during the pre-index period			
	Overall (N = 3447)	0 (n = 1750)	1 (n = 764)	2 to 5 (n = 761)	≥6 (n = 172)
Age (in years) on SCZ diagnosis date					
Mean (SD)	22.3 (2.0)	22.4 (2.0)	22.2 (2.0)	22.2 (2.0)	22.2 (2.0)
Median (IQR)	22.0 (21.0, 24.0)	23.0 (21.0, 24.0)	22.0 (21.0, 24.0)	22.0 (21.0, 24.0)	22.0 (21.0, 24.0)
Male	2,672 (77.5%)	1,388 (79.3%)	586 (76.7%)	577 (75.8%)	121 (70.3%)
Race					
White	1,168 (33.9%)	571 (32.6%)	267 (34.9%)	260 (34.2%)	70 (40.7%)
Black	1,111 (32.2%)	521 (29.8%)	255 (33.4%)	268 (35.2%)	67 (39.0%)
Hispanic	323 (9.4%)	188 (10.7%)	55 (7.2%)	NR	NR
Other[a]	693 (20.1%)	386 (22.1%)	151 (19.8%)	135 (17.7%)	21 (12.2%)
Missing	152 (4.4%)	84 (4.8%)	36 (4.7%)	NR	NR
Urban	2,921 (84.7%)	1,468 (83.9%)	647 (84.7%)	654 (85.9%)	152 (88.4%)
Days from first observed schizophrenia diagnosis date to index date					
Mean (SD)	592.0 (686.0)	370.3 (529.1)	586.2 (645.1)	918.8 (735.4)	1,427.6 (823.9)
Median (IQR)	331.0 (92.0, 848.0)	162.0 (43.0, 461.0)	373.0 (127.0, 822.5)	719.0 (346.0, 1,280.0)	1,251.5 (759.5, 1,996.5)
Without relapses in 1-month pre-index period	2,417 (70.1%)	1,422 (81.3%)	491 (64.3%)	428 (56.2%)	76 (44.2%)
OAP agents used in pre-index period					
Any OAP use	2,460 (71.4%)	983 (56.2%)	603 (78.9%)	706 (92.8%)	168 (97.7%)
Haloperidol	683 (19.8%)	195 (11.1%)	163 (21.3%)	244 (32.1%)	81 (47.1%)
Quetiapine	674 (19.6%)	218 (12.5%)	143 (18.7%)	229 (30.1%)	84 (48.8%)
Olanzapine	871 (25.3%)	277 (15.8%)	188 (24.6%)	307 (40.3%)	99 (57.6%)
Risperidone	1,188 (34.5%)	411 (23.5%)	280 (36.6%)	394 (51.8%)	103 (59.9%)
Fluphenazine	157 (4.6%)	39 (2.2%)	36 (4.7%)	60 (7.9%)	22 (12.8%)
Aripiprazole	756 (21.9%)	282 (16.1%)	187 (24.5%)	214 (28.1%)	73 (42.4%)
Clozapine	146 (4.2%)	40 (2.3%)	31 (4.1%)	52 (6.8%)	23 (13.4%)
Ziprasidone	299 (8.7%)	101 (5.8%)	56 (7.3%)	100 (13.1%)	42 (24.4%)
Paliperidone	371 (10.8%)	148 (8.5%)	78 (10.2%)	118 (15.5%)	27 (15.7%)
Lurasidone	288 (8.4%)	93 (5.3%)	57 (7.5%)	106 (13.9%)	32 (18.6%)
Any other agent	30 (0.9%)	18 (1.0%)	NR	NR	NR
Evidence of mental health-related hospitalization in 30-day pre-index period	680 (19.7%)	61 (3.5%)	209 (27.4%)	316 (41.5%)	94 (54.7%)

IQR, interquartile range; OAP, oral antipsychotic; SCZ, schizophrenia.
[a]Non-White, non-Black, non-Hispanic.

Table 3: Agent of first observed LAI use

		Number of schizophrenia-related hospitalizations during the pre-index period			
	Overall (N = 3447)	0 (n = 1750)	1 (n = 764)	2 to 5 (n = 761)	≥6 (n = 172)
FGA LAI					
Fluphenazine	170 (4.9%)	82 (4.7%)	NR	40 (5.3%)	NR
Haloperidol	597 (17.3%)	253 (14.5%)	150 (19.6%)	157 (20.6%)	37 (21.5%)
SGA LAI					
Aripiprazole	550 (16.0%)	299 (17.1%)	120 (15.7%)	106 (13.9%)	25 (14.5%)
Olanzapine	45 (1.3%)	19 (1.1%)	NR	11 (1.4%)	NR
Paliperidone	1,590 (46.1%)	830 (47.4%)	350 (45.8%)	337 (44.3%)	73 (42.4%)
Risperidone	495 (14.4%)	267 (15.3%)	96 (12.6%)	110 (14.5%)	22 (12.8%)

FGA, first-generation antipsychotics; SGA, second-generation antipsychotics; IQR, interquartile range; LAI, long-acting injectable.
NR: Value cannot be reported per the cell size suppression policy of CMS because the cell size is <11 patients or would permit calculation of a cell with <11 patients.

Table 4: Descriptive outcomes

		Number of schizophrenia-related hospitalizations during the pre-index period			
	Overall (N = 3447)	0 (n = 1750)	1 (n = 764)	2 to 5 (n = 761)	≥6 (n = 172)
Adherence (PDC≥0.80)					
To any LAI	1,165 (33.8%)	618 (35.3%)	266 (34.8%)	244 (32.1%)	37 (21.5%)
To any antipsychotics	1,890 (54.8%)	964 (55.1%)	426 (55.8%)	419 (55.1%)	81 (47.1%)
Discontinuation (gap of ≥60 days)					
Of any LAI	1,916 (55.6%)	934 (53.4%)	418 (54.7%)	442 (58.1%)	122 (70.9%)
Of any antipsychotic	1,227 (35.6%)	600 (34.3%)	284 (37.2%)	266 (35.0%)	77 (44.8%)
Evidence of relapse	1,820 (52.8%)	852 (48.7%)	388 (50.8%)	454 (59.7%)	126 (73.3%)
Hospitalization					
All-cause hospitalization	1,316 (38.2%)	491 (28.1%)	303 (39.7%)	399 (52.4%)	123 (71.5%)
Mental-health related hospitalization	1,253 (36.4%)	457 (26.1%)	282 (36.9%)	392 (51.5%)	122 (70.9%)
Schizophrenia-related hospitalization	1,101 (31.9%)	388 (22.2%)	240 (31.4%)	356 (46.8%)	117 (68.0%)

LAI, long-acting injectable; PDC, proportion of days covered.

Table 5: Logistic regression results for outcomes over 12 months following LAI initiation

	0	Number of schizophrenia-related hospitalizations during the pre-index period		
	OR (95% CI)	1 OR (95% CI)	2 to 5 OR (95% CI)	≥6 OR (95% CI)
Adherence (PDC ≥0.80)				
To any LAI	REF	1.02 (0.84, 1.24)	0.89 (0.71, 1.11)	0.49 (0.32, 0.76)
To any antipsychotics	REF	1.00 (0.83, 1.21)	0.86 (0.69, 1.06)	0.49 (0.34, 0.72)
Discontinuation (gap of ≥60 days)				
Of any LAI	REF	0.99 (0.82, 1.19)	1.13 (0.92, 1.40)	2.01 (1.35, 2.98)
Of any antipsychotic	REF	1.15 (0.95, 1.40)	1.19 (0.95, 1.48)	2.18 (1.48, 3.21)
Evidence of relapse	REF	1.01 (0.83, 1.23)	1.47 (1.18, 1.84)	2.73 (1.80, 4.16)
Any hospitalization				
All-cause hospitalization	REF	1.38 (1.13, 1.67)	2.03 (1.63, 2.52)	3.92 (2.62, 5.86)
Mental-health related hospitalization	REF	1.34 (1.10, 1.64)	2.16 (1.73, 2.68)	4.20 (2.81, 6.29)
Schizophrenia-related hospitalization	REF	1.31 (1.06, 1.61)	2.24 (1.79, 2.80)	4.93 (3.31, 7.36)

LAI, long-acting injectable; OR, odds ratio; PDC, proportion of days covered.

Key Takeaway

Greater hospitalization burden prior to LAI initiation was associated with worse subsequent adherence and higher risks of relapse and hospitalization

Conclusions

Among young dual-eligible beneficiaries with SCZ, greater hospitalization burden prior to LAI initiation was associated with worse subsequent adherence and higher risks of relapse and hospitalization

Initiating LAIs earlier may represent a critical opportunity to improve adherence, stabilize symptoms, and reduce healthcare utilization

Acknowledgements

The authors thank Elizabeth Daloisio (ApotheCom, Yardley, PA) for assistance in the preparation of this poster, which was funded by Johnson & Johnson.

Disclosures

PL reports receiving personal fees from SKB Consulting Inc and Cobbs Creek Healthcare, all unrelated to the submitted work. CB is employed by Johnson & Johnson, Titusville, NJ, and a stockholder in Johnson and Johnson. JD reports receiving grants from Johnson & Johnson, Titusville, NJ, during the conduct of the study; personal fees from AbbVie, Acadia, Janssen, Merck, Otsuka, and Takeda; and grants from Merck and Spark Therapeutics unrelated to the submitted work. No other authors had disclosures to report. This study was funded by Johnson & Johnson.

Neuropsychiatry

Scan the QR code

The QR code is intended to provide scientific information for individual reference, and the information should not be altered or reproduced in any way.

This study was funded by Johnson & Johnson.

Presented at Psych Congress; September 15-19, 2026; New Orleans, Louisiana, USA

References

1. American Psychiatric Association. *Diagnostic and Statistical Manual of Mental Disorders: DSM-5TM, 5th Ed.* American Psychiatric Publishing, Inc.; 2013. 2. Marcus SC, Zummo J, Pettit AR, Stoddard J, Doshi JA. Antipsychotic Adherence and Rehospitalization in Schizophrenia Patients Receiving Oral Versus Long-Acting Injectable Antipsychotics Following Hospital Discharge. *J Manag Care Spec Pharm.* 2015;21(9):754-768. 3. El Abdellati K, De Picker L, Morrens M. Antipsychotic Treatment Failure: A Systematic Review on Risk Factors and Interventions for Treatment Adherence in Psychosis. *Front Neurosci.* 2020;14:531763. doi:10.3389/fnins.2020.531763 4. Olivares JM, Sermon J, Hemels M, Schreiner A. Definitions and drivers of relapse in patients with schizophrenia: a systematic literature review. *Ann Gen Psychiatry.* 2013;12(1). Accessed July 30, 2026. https://pubmed.ncbi.nlm.nih.gov/24148707/ 5. Lin D, Thompson-Leduc P, Ghelerter I, et al. Real-World Evidence of the Clinical and Economic Impact of Long-Acting Injectable Versus Oral Antipsychotics Among Patients with Schizophrenia in the United States: A Systematic Review and Meta-Analysis. *CNS Drugs.* 2021;35(5):469-481. doi:10.1007/s40263-021-00815-y 6. Li P, Geng Z, Benson C, Patel C, Doshi JA. Real-World Effectiveness of Long-Acting Injectable and Oral Antipsychotic Agents in US Medicare Patients with Schizophrenia. *Adv Ther.* 2025;42(2):1251-1264. doi:10.1007/s12325-024-03075-6 7. Kane JM, Mychaskiw MA, Lim S, Suett M, Tian M, Rubio JM. Treatment Journey From Diagnosis to the Successful Implementation of a Long-Acting Injectable Antipsychotic Agent in Young Adults With Schizophrenia. *J Clin Psychiatry.* 2023;84(3):22m14544. doi:10.4088/JCP.22m14544