106

Association of Major Depressive Disorder With Insomnia Symptoms With Healthcare Resource Use and Cardiovascular and Metabolic Conditions – Analysis of the National Health & Nutrition Examination Survey

Hrishikesh Kale[1], Zhiwen Liu[1], Karen Johnston[1], Guo Li[2], Carmela Benson[1], Zhiheng Zhang[2]

[1]Johnson & Johnson, Titusville, NJ; [2]Johnson & Johnson, Raritan, NJ

Introduction

- Major depressive disorder (MDD) is a prevalent mental health condition with a prevalence of 8.2% (21.4 million adults) in the US[1]
- MDD is a heterogeneous condition with varying symptoms such as depressed mood, anhedonia, poor appetite, concentration difficulties and sleep disturbances[1,2]
- Insomnia is one of the key symptoms of MDD and worsening of depressed mood exacerbates insomnia symptoms and vice versa[2,3]
- Previous research has found that MDD with insomnia symptoms (MDDIS) is associated with increased risk of relapse, poor health-related quality of life and greater economic burden[4-6]
- Cardiovascular and metabolic conditions are common co-existing conditions in patients with MDD; however, little is known about the impact of MDDIS on the prevalence and worsening of these comorbidities in patients with MDD

Objective

- To examine the association of MDDIS with cardiovascular and metabolic (CVM) conditions and healthcare resource utilization using nationally representative data from the National Health and Nutrition Examination Survey (NHANES)

Methods

Data source

- This study analyzed data from the NHANES database (2013-2014, 2015-2016, 2017-2018), which is a publicly available survey of the noninstitutionalized civilian US population
- The NHANES sample is selected using a complex, 4-stage sample design and the sample weights can be used to derive national estimates

Study design

- This was a cross-sectional study of adults identified from the NHANES database
- The study considered survey questions that were administered during the 3 survey cycles mentioned above
- Patients with MDD were identified using a 9-item Patient Health Questionnaire (PHQ-9) total score ≥10 (score range 0–27)
- The PHQ-9 instrument asks participants about the frequency of depressive symptoms during the previous 2 weeks
- Response categories for PHQ-9 include the following
 - 0: "not at all"
 - 1: "several days"
 - 2: "more than half the days"
 - 3: "nearly every day"
- Insomnia symptoms were identified based on self-report of trouble sleeping, using the question: "Have you ever told a doctor or other health professional that you have trouble sleeping?"
- The study sample was selected based on the following inclusion criteria:
 - Patients aged ≥18 years
 - Patients having nonmissing data for both:
 - PHQ-9 score
 - Self-report of trouble sleeping
- Among patients with MDD,
 - MDDIS was defined as having MDD and answering "Yes" to the 'trouble sleeping' question
 - MDD w/o IS was defined as having MDD and answering "No" to the 'trouble sleeping' question

Study outcomes

- Healthcare resource utilization included inpatient stays, outpatient office visits (all-cause) and mental health professional visits
 - Inpatient use was defined as an overnight hospital stay in the past year
 - Outpatient visits were defined as the number of times patients received healthcare in an outpatient/office-based setting
 - Mental health professional visit was defined as presence of any mental health professional visit in the past year
- Work productivity limitation was defined as the patient being unable to work during the past week due to health reasons
- CVM conditions were defined as below
 - Diabetes was defined as either a self-reported diagnosis by a physician (ever told by a doctor that you have diabetes), plasma hemoglobin A1c (HbA1c) ≥6.5%, or fasting plasma glucose ≥7.0 mmol/L
 - Prediabetes was defined as the presence of any of the following criteria: 1) had been told by a doctor that you have prediabetes; 2) HbA1C between 5.7% and 6.4%; 3) fasting blood glucose between 5.6 to 6.9 mmol/L in subjects without the defined diabetes
 - Hypertension was defined as systolic blood pressure (SBP) ≥130 mm Hg, diastolic blood pressure (DBP) ≥80 mm Hg, self-reported diagnosis by a physician (had been told by a doctor that you have hypertension on 2 or more different visits), or self-reported prescriptions for hypertension
 - Presence of coronary heart disease and stroke was reported if the doctor or healthcare professional ever told the patient that they had those conditions

Statistical analysis

- MDDIS and MDD w/o IS were balanced using propensity score–based inverse probability of treatment weighting (IPTW), controlling for baseline demographics
- Study outcomes were compared between MDDIS and MDD w/o IS using descriptive statistics and weighted (IPTW and sample weights) logistic regression models
- The findings reported here include sample counts and weighted percentages

Table 1: Demographics of study participants with MDD

	MDDIS		MDD w/o IS		
	N	%	N	%	P value
Sex	853		530		
Men	308	35.4	207	37.1	0.6040
Women	545	64.6	323	62.9	
Age					
Mean (SD)	49.0 (0.8)		43.3 (1.2)		<0.0001
18-24	67	7.9	94	22.0	0.0002
25-44	228	32.1	158	35.5	
45-64	375	42.4	162	25.3	
65+	183	17.7	116	17.1	
Race					
Mexican American	110	6.4	97	11.6	<0.0001
Other Hispanic	102	6.3	74	9.3	
Non-Hispanic White	384	68.3	178	53.5	
Non-Hispanic Black	168	10.1	120	15.3	
Non-Hispanic Asian	29	1.6	33	3.4	
Other	60	7.2	28	7.0	
Marital status					
Married	308	39.5	159	29.8	0.0040
Not married	517	60.5	337	70.2	
Education					
Less than high school	243	19.2	174	26.0	0.0228
High school	485	63.8	262	60.6	
Above college	98	17.1	58	13.4	

MDDIS, major depressive disorder with insomnia symptoms; MDD w/o IS, major depressive disorder without insomnia symptoms. Data are expressed as the unweighted number of participants and survey-weighted percentages to be nationally representative. Data represents frequency and percentages before inverse probability of treatment weighting (IPTW).

Results

Patient characteristics

- From a sample of 15,574 adults, 1383 (8.2%) had MDD (PHQ-9 ≥10) and 853 (5.4%) had MDDIS
- Of 1383 patients with MDD, 853 (66%) were categorized as having MDDIS and 530 (34%) were categorized as having MDD w/o IS
- Compared with patients with MDD w/o IS, patients with MDDIS were older (mean, 49.0 vs 43.3 years), White (68.3% vs 53.5%), married (39.5% vs 29.8%) and college educated (17.1% vs 13.4%) (**Table 1**)
- The proportion of females was similar between those with MDDIS (64.6%) and MDD w/o IS (62.9%)

Cardiovascular and metabolic conditions

- Patients with MDDIS versus MDD w/o IS had significantly higher (*P* < 0.05) odds for heart attack (7.4% vs 4.3%; odds ratio [OR]: 1.76) and hypertension (64.7% vs 46.6%; OR: 2.10) (**Figure 1A**)
- Findings for obesity (54.4% vs 44.4%; OR: 1.50) and diabetes (21.7% vs 17.6%; OR: 1.29) were not statistically significant but showed a similar trend (**Figure 1B**)

Figure 1: Associations between MDDIS and cardiovascular and metabolic conditions in patients with MDD

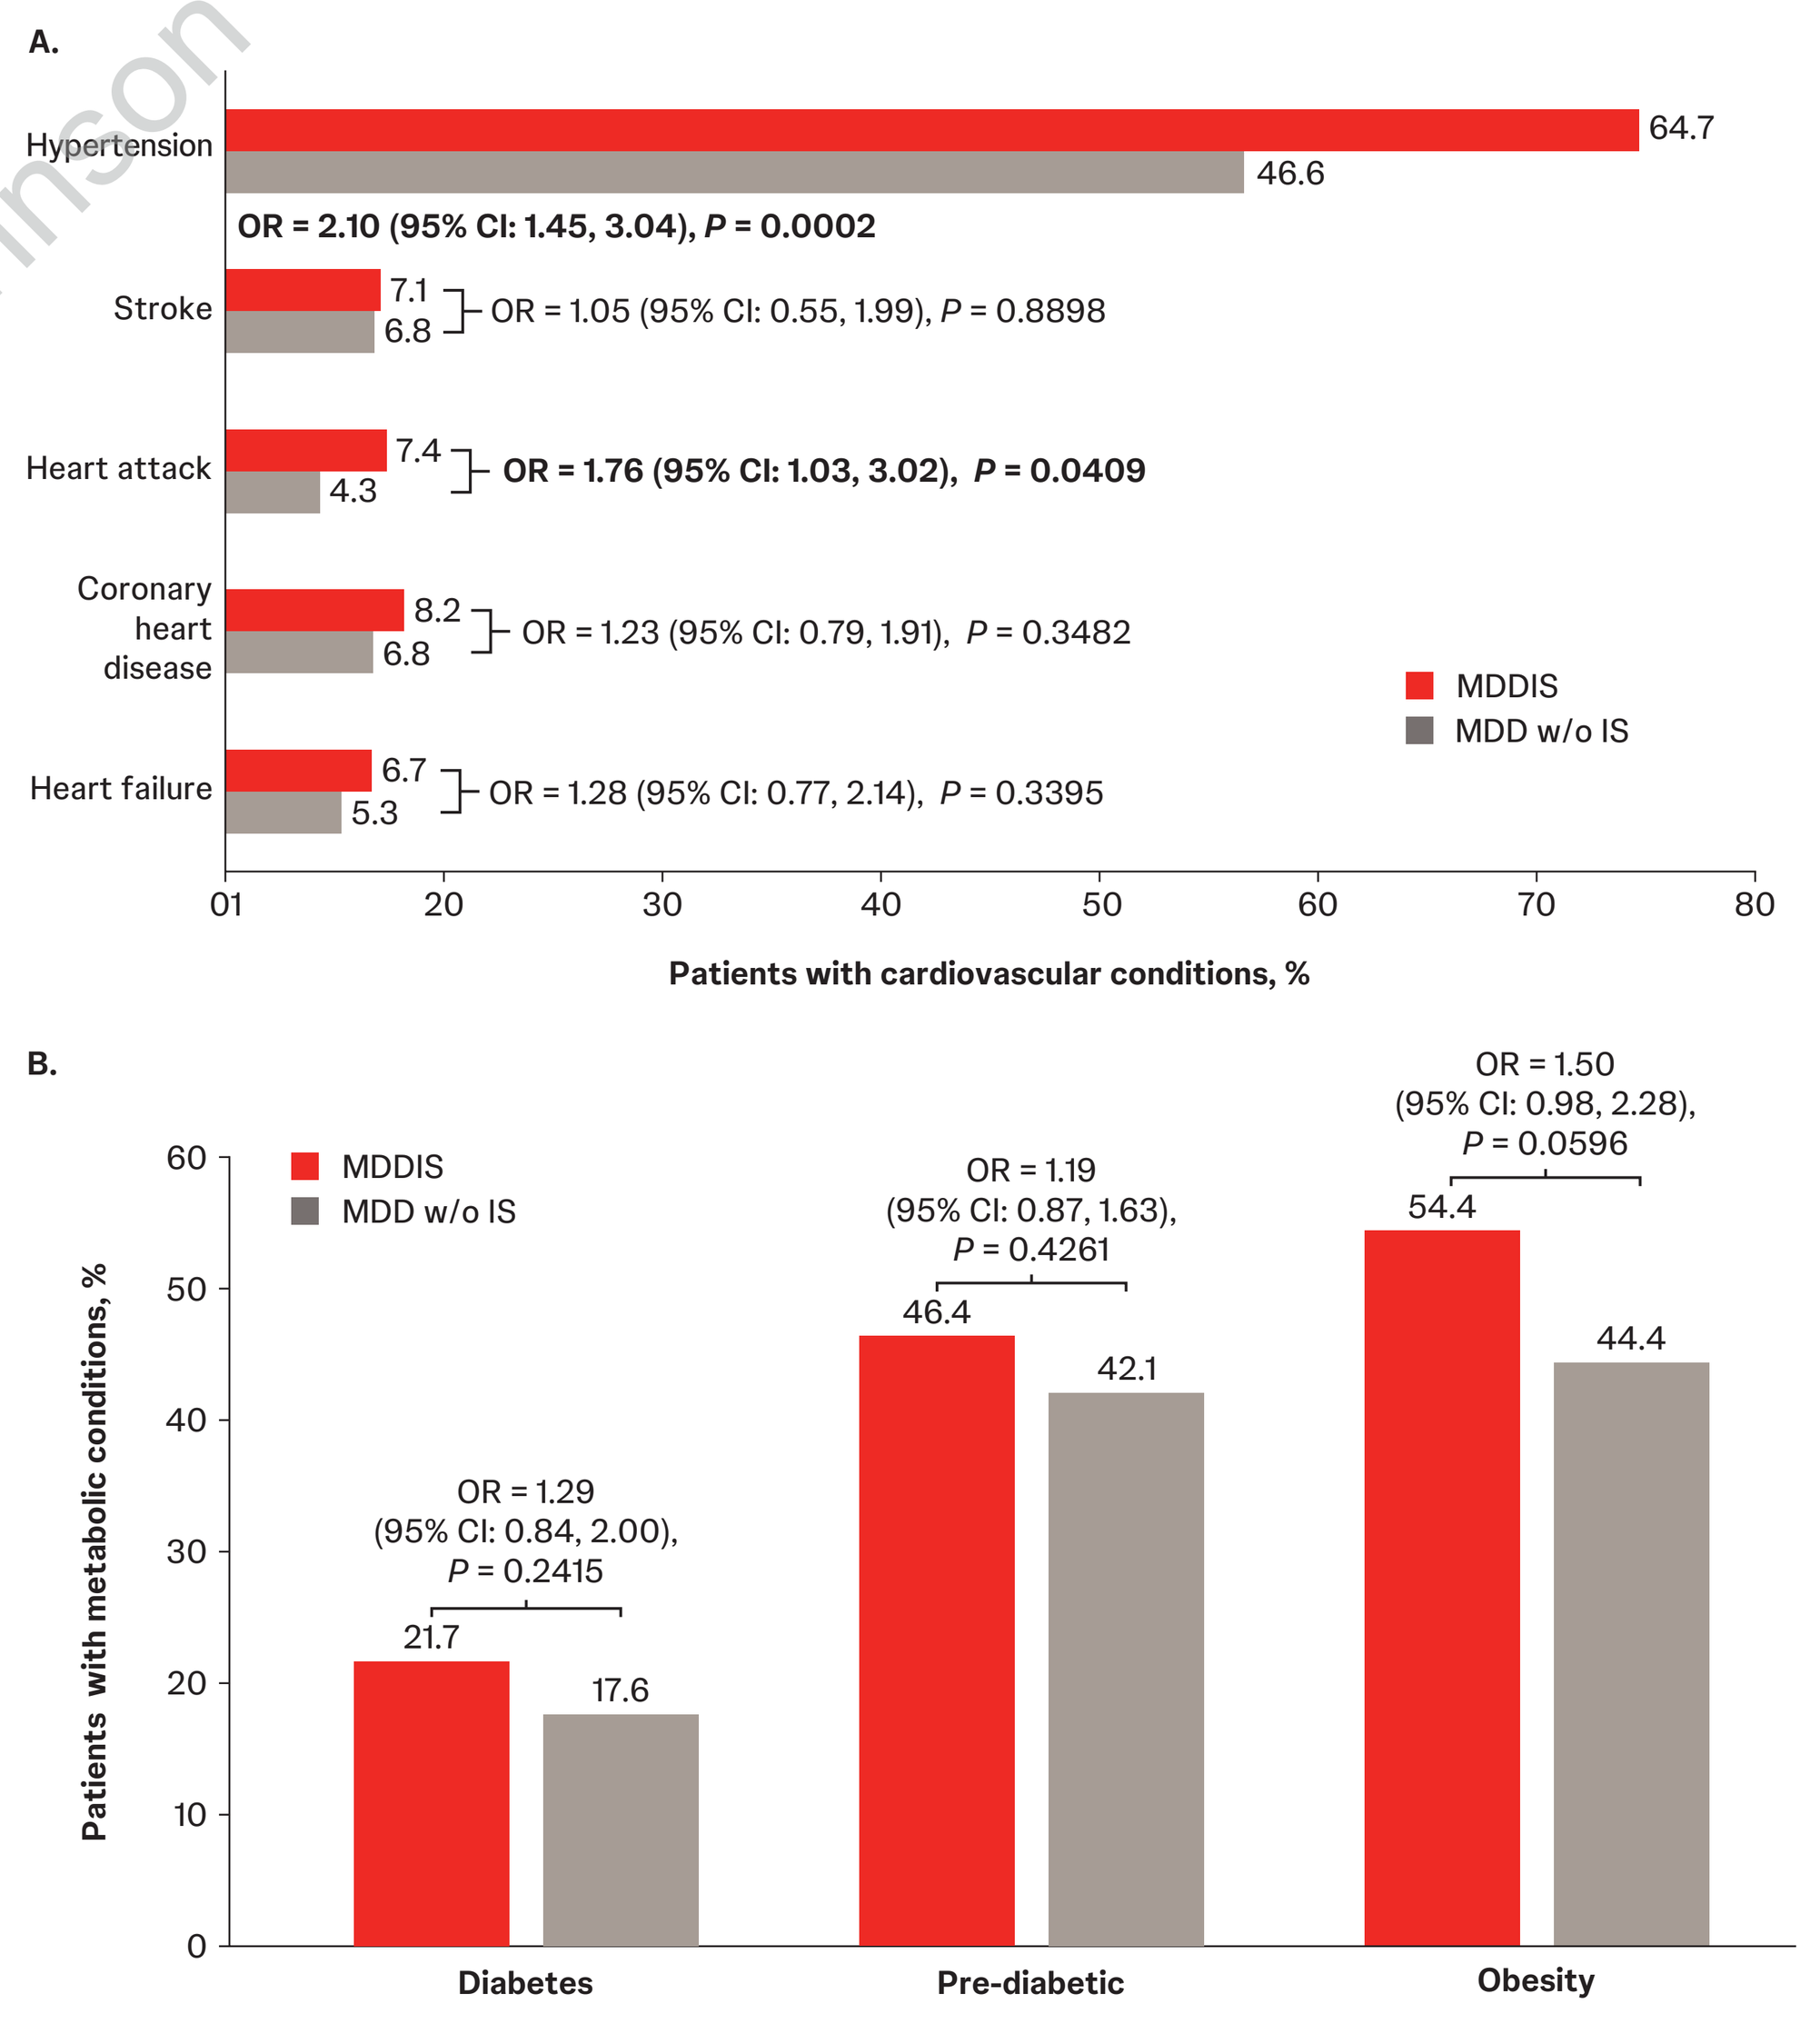
A.
Hypertension
64.7
46.6
OR = 2.10 (95% CI: 1.45, 3.04), P = 0.0002
Stroke
7.1
6.8
OR = 1.05 (95% CI: 0.55, 1.99), P = 0.8898
Heart attack
7.4
4.3
OR = 1.76 (95% CI: 1.03, 3.02), P = 0.0409
Coronary heart disease
8.2
6.8
OR = 1.23 (95% CI: 0.79, 1.91), P = 0.3482
Heart failure
6.7
5.3
OR = 1.28 (95% CI: 0.77, 2.14), P = 0.3395
MDDIS
MDD w/o IS
01
20
30
40
50
60
70
80
Patients with cardiovascular conditions, %
B.
Patients with metabolic conditions, %
0
10
20
30
40
50
60
MDDIS
MDD w/o IS
OR = 1.29
(95% CI: 0.84, 2.00),
P = 0.2415
21.7
17.6
OR = 1.19
(95% CI: 0.87, 1.63),
P = 0.4261
46.4
42.1
OR = 1.50
(95% CI: 0.98, 2.28),
P = 0.0596
54.4
44.4
Diabetes
Pre-diabetic
Obesity

MDD, major depressive disorder; MDDIS, major depressive disorder with insomnia symptoms; MDD w/o IS, major depressive disorder without insomnia symptoms; OR, odds ratio.

Figure 2: Associations between MDDIS and healthcare resource utilization and work productivity in patients with MDD

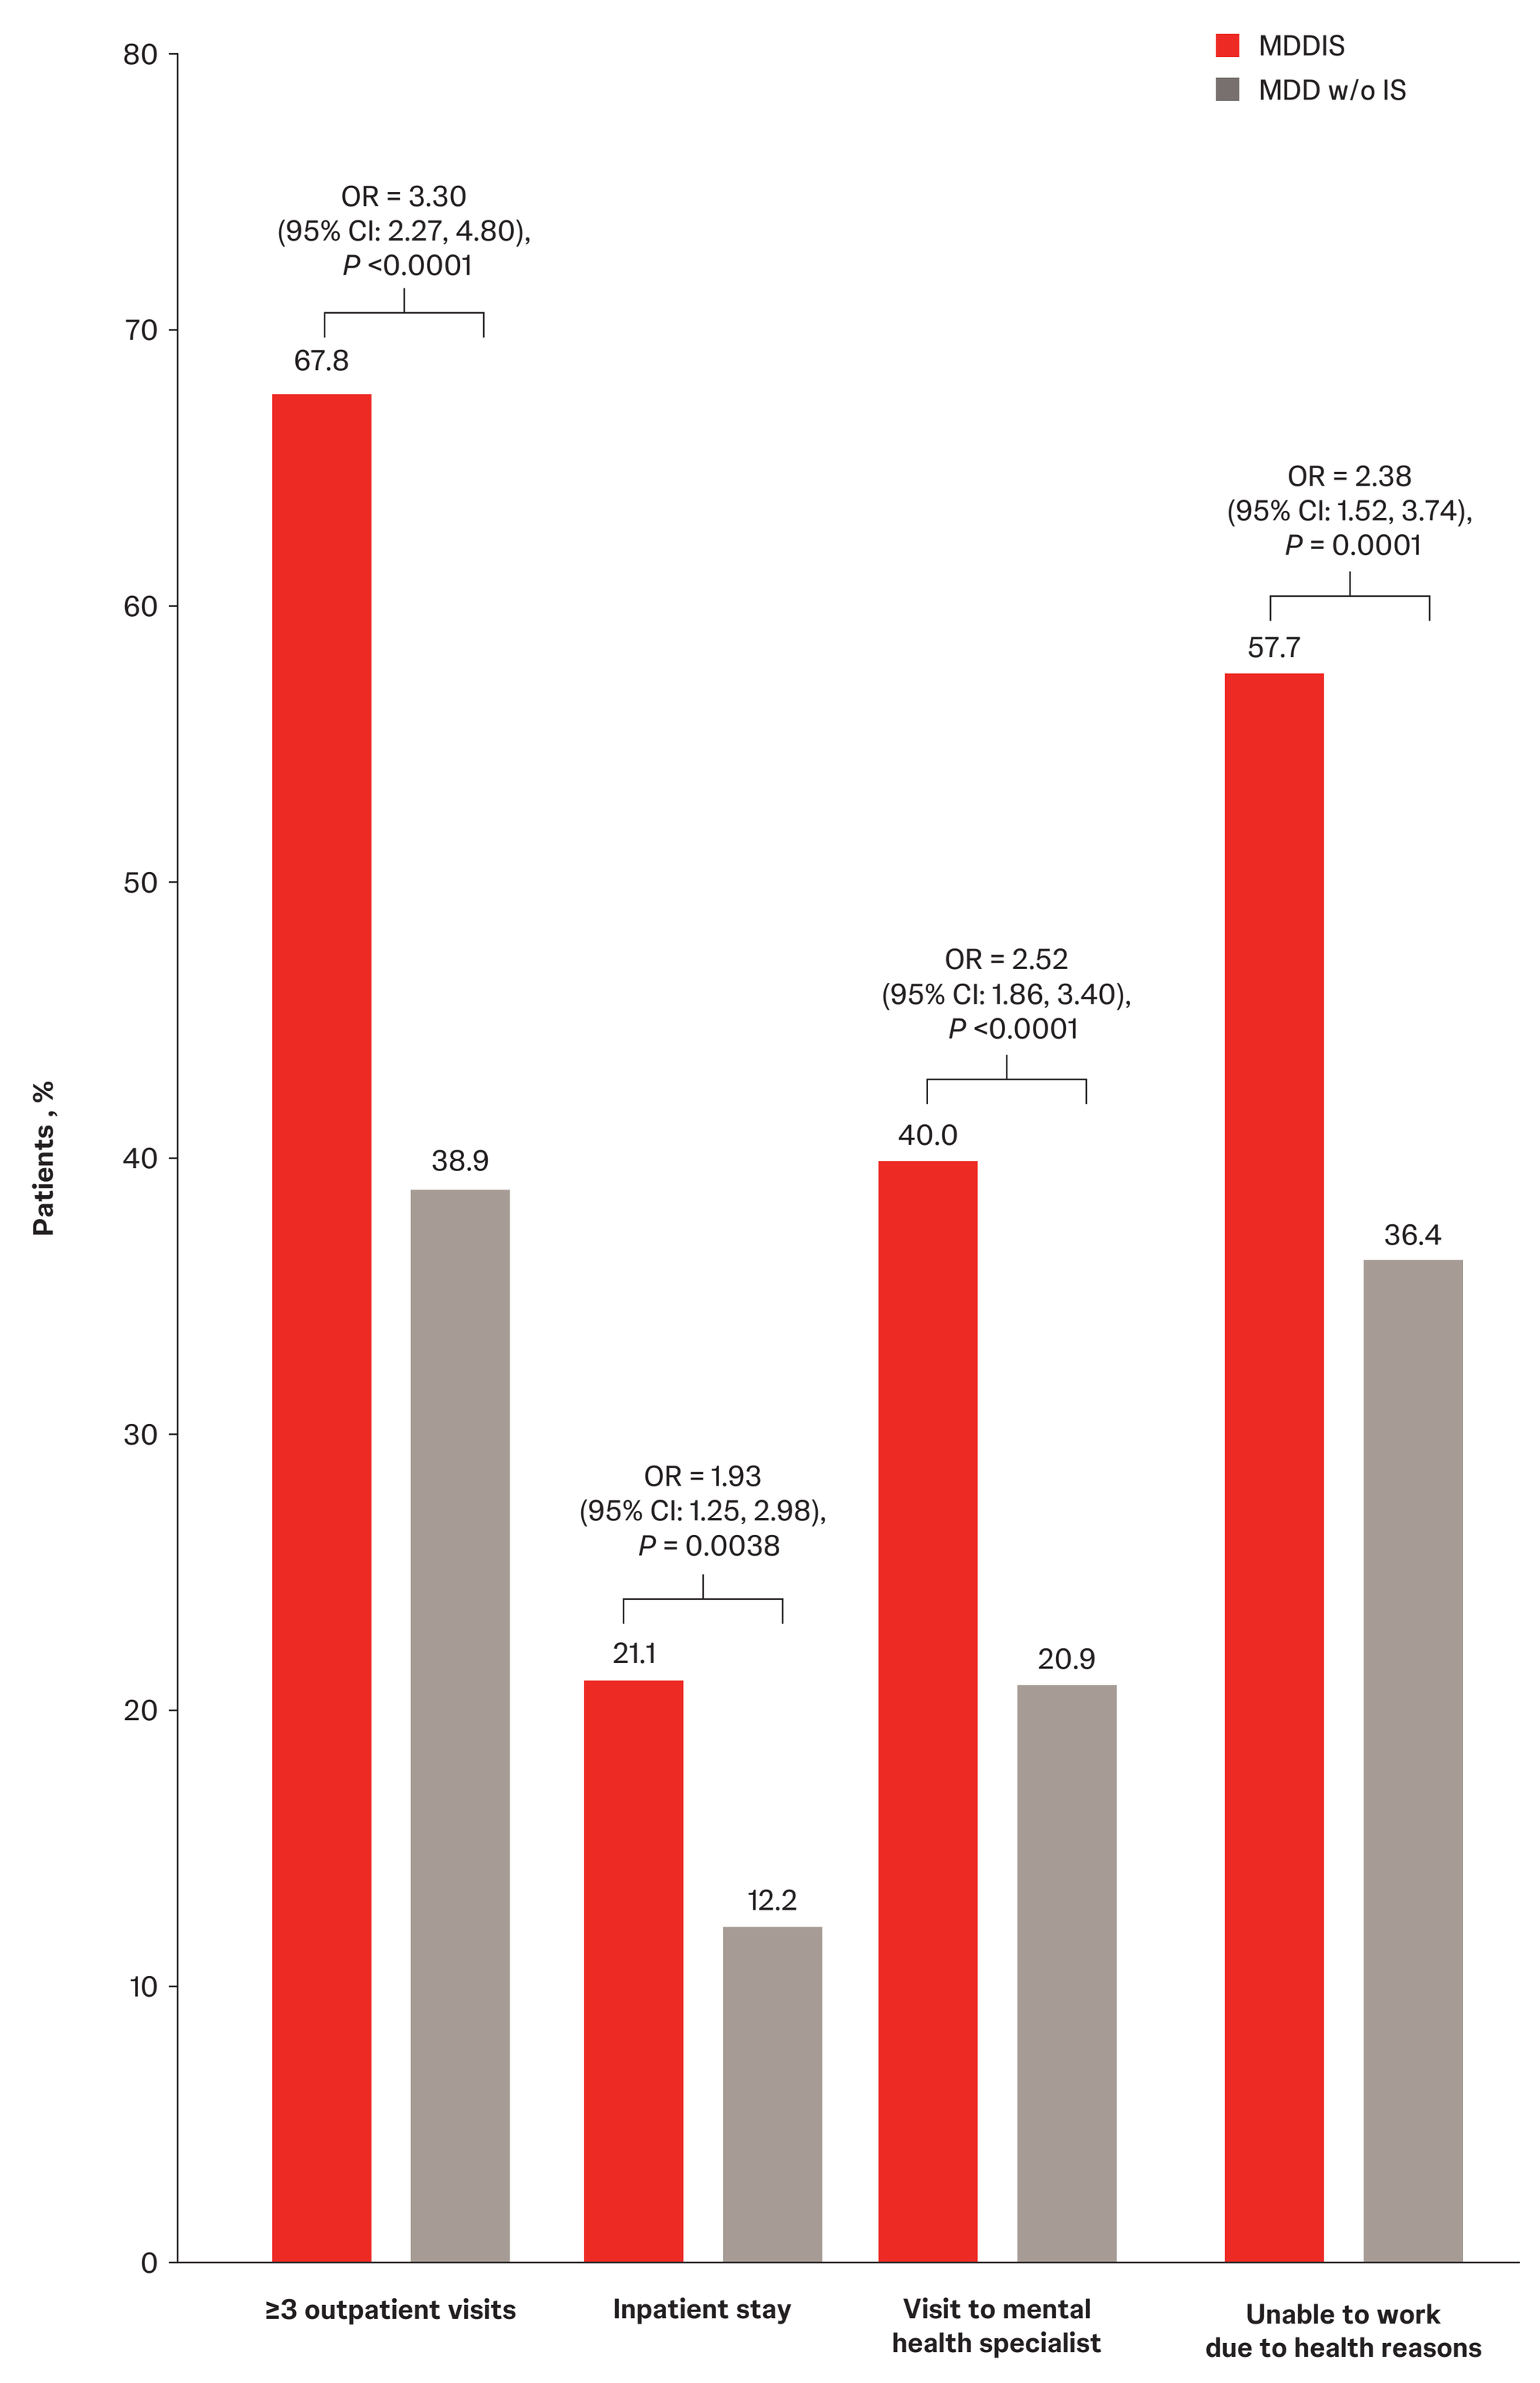
MDDIS
MDD w/o IS
Patients , %
0
10
20
30
40
50
60
70
80
OR = 3.30
(95% CI: 2.27, 4.80),
P <0.0001
67.8
38.9
OR = 1.93
(95% CI: 1.25, 2.98),
P = 0.0038
21.1
12.2
OR = 2.52
(95% CI: 1.86, 3.40),
P <0.0001
40.0
20.9
OR = 2.38
(95% CI: 1.52, 3.74),
P = 0.0001
57.7
36.4
≥3 outpatient visits
Inpatient stay
Visit to mental health specialist
Unable to work due to health reasons

MDD, major depressive disorder; MDDIS, major depressive disorder with insomnia symptoms; MDD w/o IS, major depressive disorder without insomnia symptoms; OR, odds ratio.

Healthcare resource utilization and work productivity

- Results from weighted logistic regression analyses (**Figure 2**) found that MDDIS patients had significantly (*P* < 0.05) higher odds than those with MDD w/o IS for
 - ≥3 office visits (67.8% vs 38.9%; OR: 3.30)
 - Hospitalization (21.1% vs 12.2%; OR: 1.93)
 - Mental health professional visit (40.0% vs 20.9%; OR: 2.52)
 - Inability to work due to health reasons (57.7% vs 36.4%; OR: 2.38)

Key Takeaway

There remains a critical need for therapies that effectively address both depressive and insomnia symptoms in patients with MDD

Limitations

- NHANES is a cross-sectional database; therefore, only associations can be derived between presence of insomnia symptoms in MDD and outcomes; causality cannot be implied
- Current findings do not distinguish between prevalent versus incident cardiovascular and metabolic conditions
- The study did not assess depression treatment use due to lack of prescription medication data in NHANES
- Study findings are subject to respondent recall and response bias due to the self-reported nature of the survey questionnaire data
- Findings from this study may not be generalizable to institutionalized or nursing home populations

Conclusions

- MDDIS was associated with higher healthcare resource utilization, reduced work productivity, and poorer CVM health
- Study findings indicate that the burden of MDDIS extends beyond mental health, contributing to broader clinical and societal impacts
- There remains a critical need for therapies that effectively address both depressive and insomnia symptoms in patients with MDD

Acknowledgments

The authors thank Alicia K Campbell, PharmD, for her assistance in writing the abstract and the study protocol. Editorial assistance was provided by ApotheCom, Yardley, PA, which was funded by Johnson & Johnson.

Disclosures

HK, ZL, KJ, GL, CB, and ZZ are employees of Johnson & Johnson and hold stock in the company.

Funding

This study was funded by a Johnson & Johnson company.

Previously presented at the American Society of Clinical Psychopharmacology (ASCP) Annual Meeting; May 26-29, 2026; Miami, Florida, USA.

Novel Pathways in Depression

Scan the QR code

The QR code is intended to provide scientific information for individual reference, and the information should not be altered or reproduced in any way.

Presented at Psych Congress; September 15-19, 2026; New Orleans, Louisiana, USA

References

1. Substance Abuse and Mental Health Services Administration. Key substance use and mental health indicators in the United States: Results from the 2024 National Survey on Drug Use and Health. Publication Number PEP25-07-007, NSDUH Series H-60. 2025. 2. Peterson MJ et al. *Sleep Med Clin*. 2008;3(2):231-249. 3. Sivertsen B et al. *Biopsychosocial Sci Med*. 2012;74(7):758-765. 4. Sakurai H et al. *Psychopharmacology*. 2017;234(16):2453-2461. 5. Joshi K et al. *J Affect Disord*. 2023;323:698-706. 6. Kale H et al. *J Affect Disord*. 2025;372:653-664.

This material is distributed for scientific purposes by Johnson & Johnson Innovative Medicine and is not for promotional use.