4910

Long-Term Safety and Efficacy of Esketamine Nasal Spray Among Hispanic and Latino Adults With Treatment-Resistant Depression in the SUSTAIN-3 Study

Lisa Harding[1], Ibrahim Turkoz[2], Larry Martinez[2], Clairissa Cruz[2]

[1]Depression MD, Milford, CT; [2]Johnson & Johnson, Titusville, NJ

Introduction

- Hispanic and Latino patients with depression experience barriers to mental healthcare and treatment inequalities compared with White patients[1-3]
 - In the 2021 National Survey on Drug Use and Health, 36.1% of Hispanic adults compared with 52.4% of White adults received mental health services for mental illness in the United States (US), and 41.8% of Hispanic adults compared with 58.3% of White adults took prescription medication for mental health issues[1]
 - In the US, Hispanic and Latino patients face barriers to mental healthcare, including language barriers, poverty, less health insurance coverage, lack of cultural competence of the provider, legal status issues, low level of acculturation, and stigma regarding discussing and receiving treatment for mental illness[2]
 - Additionally, White patients may be more likely to be treated with new medications compared with Hispanic patients[3]
- These disparities highlight the importance of understanding the efficacy and safety of antidepressant therapies in the Hispanic and Latino populations
- Esketamine nasal spray (ESK) is approved by the US Food and Drug Administration for the treatment of treatment-resistant depression (TRD) in adults, as monotherapy or in conjunction with an oral antidepressant (OAD), and for the treatment of depressive symptoms in adults with major depressive disorder with acute suicidal ideation or behavior in conjunction with an OAD[4]
- SUSTAIN-3 was an open-label extension study of flexibly dosed ESK, in conjunction with an OAD, which lasted up to 6.5 years and demonstrated that improvements in depression generally persisted among patients who remained on maintenance treatment; no new safety signals were identified with long-term treatment[5,6]

Objective

- This subgroup analysis of SUSTAIN-3 evaluated the long-term safety and efficacy of flexibly dosed ESK, in combination with an OAD, among Hispanic and Latino adults with TRD

Methods

Study design

- SUSTAIN-3 (NCT02782104) was a multicenter, open-label, long-term extension study; patients entered SUSTAIN-3 from 1 of 6 parent studies (**Figure 1**)
- Patients enrolled into a 4-week induction (IND) phase or an optimization/maintenance (OP/M) phase if they had received IND in a previous study
- Patients received flexibly dosed ESK (56 or 84 mg) in conjunction with an OAD
- This subgroup analysis included Hispanic or Latino patients aged 18-64 years

Data analyses

- The clinician-rated Montgomery-Åsberg Depression Rating Scale (MADRS; range 0-60) and the patient-reported Patient Health Questionnaire-9 (PHQ-9; range 0-27) were used to evaluate depression symptom severity over time, with higher scores indicating more severe depression
- Response rates (defined as a ≥50% improvement in the MADRS or PHQ-9 total score) were measured for patients of the IND phase
- Remission rates (defined as an MADRS total score ≤12 or PHQ-9 total score <5) were measured for patients of the IND and OP/M phases
- The patient-reported Sheehan Disability Scale (SDS; range 0-30) was used to measure functional impairment in the domains of work/school, social life, and family life/home responsibilities over time, with a higher score indicating greater impairment
- Treatment-emergent adverse events (TEAEs) were monitored throughout the study

Figure 1: SUSTAIN-3 study design

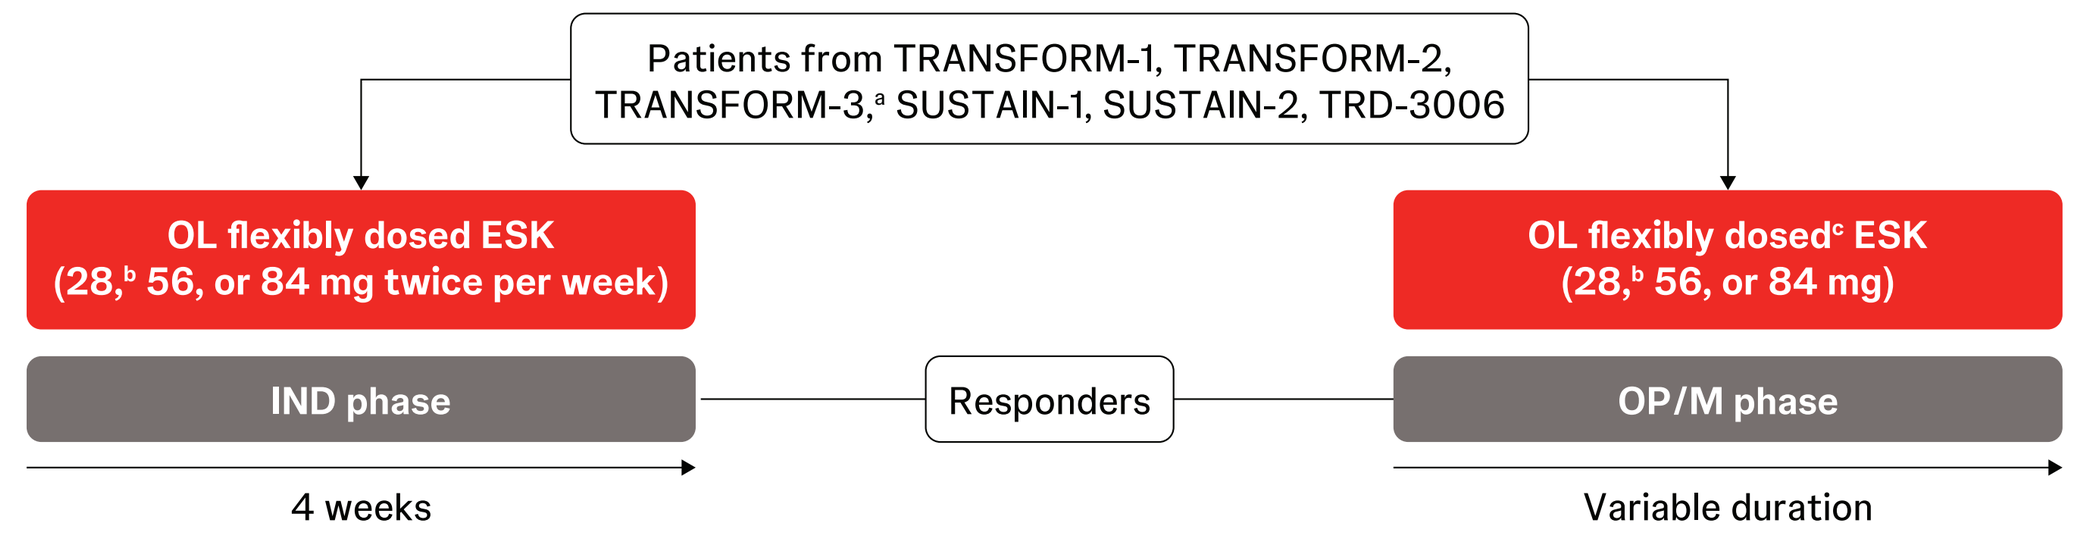
Patients from TRANSFORM-1, TRANSFORM-2, TRANSFORM-3,[a] SUSTAIN-1, SUSTAIN-2, TRD-3006
OL flexibly dosed ESK (28,[b] 56, or 84 mg twice per week)
OL flexibly dosed[c] ESK (28,[b] 56, or 84 mg)
IND phase
Responders
OP/M phase
4 weeks
Variable duration

ESK, esketamine nasal spray; IND, induction; OL, open label; OP/M, optimization/maintenance.
[a]TRANSFORM-3 patients (aged ≥65 years) were not included in this analysis.
[b]The 28-mg dose was only an option for patients aged ≥65 years, who were not included in this analysis.
[c]Based on Clinical Global Impression-Severity scale and tolerability.

Results

Disposition, demographics, and baseline characteristics

- A total of 1021 patients between the ages of 18 to 64 years were enrolled in SUSTAIN-3, of whom 182 were Hispanic or Latino adults (**Table 1**)
- Among Hispanic and Latino patients, 34.6% (n = 63) enrolled into the IND phase and 65.4% (n = 119) enrolled into the OP/M phase
- Patients who enrolled into the IND phase had a mean (SD) MADRS total score of 28.9 (8.39) and a mean (SD) PHQ-9 total score of 14.8 (5.87), indicating moderate to severe disease

TABLE 1: Patient demographics and baseline clinical characteristics

	Hispanic/Latino n = 182	Total N = 1021
Mean age (SD), years	48.6 (10.55)	47.3 (10.71)
Female, n (%)	152 (83.5)	678 (66.4)
Race, n (%)		
White	155 (85.6)	874 (85.6)
Other/multiple races	22 (12.2)	38 (3.7)
Black or African American	3 (1.7)	44 (4.3)
Asian	0	42 (4.1)
Ethnicity, n (%)		
Hispanic or Latino	182 (100)	182 (17.8)
Mean age when diagnosed with MDD (SD), years	35.7 (11.78)	32.8 (11.86)
Mean duration of current episode (range), weeks	167.2 (6-1272)	153.3 (6-2288)
Patients entering at IND phase, n (%)	63 (34.6)	440 (43.1)
Mean baseline MADRS total score (SD)	28.9 (8.39)	29.1 (7.94)
Mean baseline PHQ-9 total score (SD)	14.8 (5.87)	15.4 (5.56)

IND, induction; MADRS, Montgomery-Åsberg Depression Rating Scale; MDD, major depressive disorder; PHQ-9, Patient Health Questionnaire-9.

Exposure

- A total of 83.0% of Hispanic and Latino patients had ≥24 months of ESK treatment, 52.2% had ≥36 months of ESK treatment, and 2.7% had ≥42 months of ESK treatment (**Figure 2**)

Safety

- Most Hispanic and Latino patients experienced ≥1 TEAE during the study (95.6%), and most TEAEs were mild or moderate in severity and were resolved on the same day as dosing
- The most common TEAEs experienced by Hispanic and Latino patients were dizziness (48.9%), headache (44.5%), somnolence (44.0%), dissociation (40.7%), and nausea (35.2%) and were the same most common TEAEs as those experienced in the overall SUSTAIN-3 patient population (**Table 2**)
- Rate of discontinuation from the study due to adverse events was 1.6% in the IND phase and 2.8% in the OP/M phase

Efficacy

- The mean MADRS and PHQ-9 total scores over time are shown in **Figure 3**; mean SDS total scores over time are shown in **Figure 4**
- The mean (SD) change in MADRS total score from baseline to IND end point was −13.6 (11.2) and the mean (SD) change in PHQ-9 total score was −5.5 (5.4)
- The mean (SD) change in MADRS total score from baseline to OP/M end point was 2.5 (9.45) and the mean (SD) change in PHQ-9 total score was 1.7 (0.47) showing that improvements seen in the IND phase were maintained in the OP/M phase
- The response rate (≥50% improvement in total score) at IND end point per MADRS total score was 52.4% and per PHQ-9 total score was 38.7% (**Table 3**)
- The proportion of patients who achieved remission at OP/M end point per MADRS total score (≤12) was 59.2% and per PHQ-9 total score (<5) was 34.1%
- The mean (SD) change in the SDS total score from baseline to IND end point was −7.2 (7.6), and the mean (SD) change in SDS total score from baseline to OP/M end point was 0.5 (0.61) , showing that improvements seen in the IND phase were maintained in the OP/M phase

TABLE 2: Most common (≥10%) TEAEs

TEAE, n (%)	Hispanic and Latino n = 182	Total N = 1021
Dizziness	89 (48.9)	354 (34.7)
Headache	81 (44.5)	382 (37.4)
Somnolence	80 (44.0)	241 (23.6)
Dissociation	74 (40.7)	270 (26.4)
Nausea	64 (35.2)	351 (34.4)
Dysgeusia	44 (24.2)	221 (21.6)
Anxiety	37 (20.3)	194 (19.0)
Influenza	36 (19.8)	126 (12.3)
Insomnia	35 (19.2)	147 (14.4)
Back pain	33 (18.1)	199 (19.5)
Urinary tract infection	32 (17.6)	152 (14.9)
Paresthesia oral	32 (17.6)	74 (7.2)
Arthralgia	29 (15.9)	164 (16.1)
Nasal discomfort	28 (15.4)	94 (9.2)
Vertigo	27 (14.8)	195 (19.1)
Paresthesia	27 (14.8)	87 (8.5)
Vomiting	26 (14.3)	166 (16.3)
Diarrhea	25 (13.7)	162 (15.9)
Hypoesthesia oral	23 (12.6)	87 (8.5)
Nasopharyngitis	22 (12.1)	235 (23.0)
Sedation	22 (12.1)	84 (8.2)
Hypoesthesia	21 (11.5)	106 (10.4)
Blood pressure increased	21 (11.5)	148 (14.5)

TEAE, treatment-emergent adverse event.

TABLE 3: Proportion of patients who achieved response and remission in the IND and OP/M phases

	Hispanic and Latino n = 182	Total N = 1021
MADRS		
IND phase end point, n/N (%)		
Response (≥50% improvement from baseline)	33/63 (52.4)	218/437 (49.9)
Remission (MADRS total score ≤12)	23/63 (36.5)	159/438 (36.3)
OP/M end point, n/N (%)		
Remission (MADRS total score ≤12)	106/179 (59.2)	489/986 (49.6)
PHQ-9		
IND phase end point, n/N (%)		
Response (≥50% improvement from baseline)	24/63 (38.7)	167/435 (38.4)
Remission (PHQ-9 total score <5)	16/63 (25.4)	88/437 (20.1)
OP/M end point, n/N (%)		
Remission (PHQ-9 total score <5)	61/179 (34.1)	331/986 (33.6)

IND, induction; MADRS, Montgomery-Åsberg Depression Rating Scale; MDD, major depressive disorder; PHQ-9, Patient Health Questionnaire-9.

FIGURE 2: ESK exposure in Hispanic and Latino patients

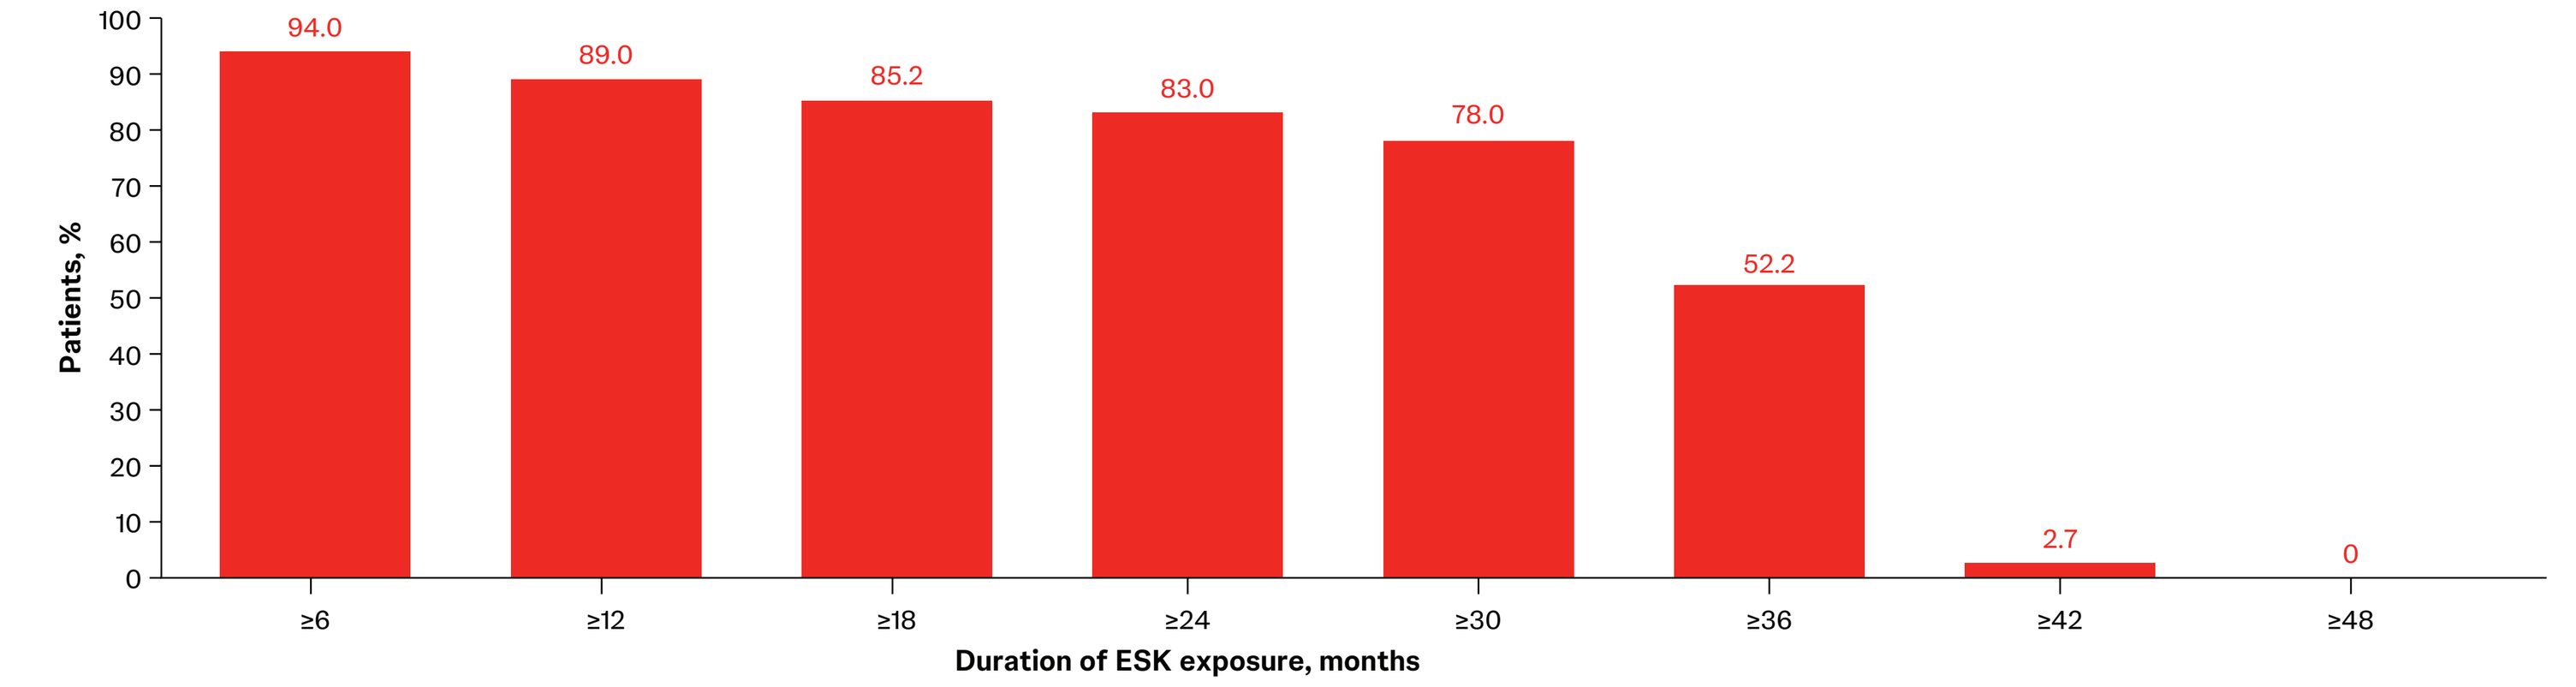
Patients, %
94.0
89.0
85.2
83.0
78.0
52.2
2.7
0
≥6
≥12
≥18
≥24
≥30
≥36
≥42
≥48
Duration of ESK exposure, months

ESK, esketamine nasal spray.

FIGURE 3: Mean (A) MADRS and (B) PHQ-9 total scores during the IND and OP/M phases (observed cases)

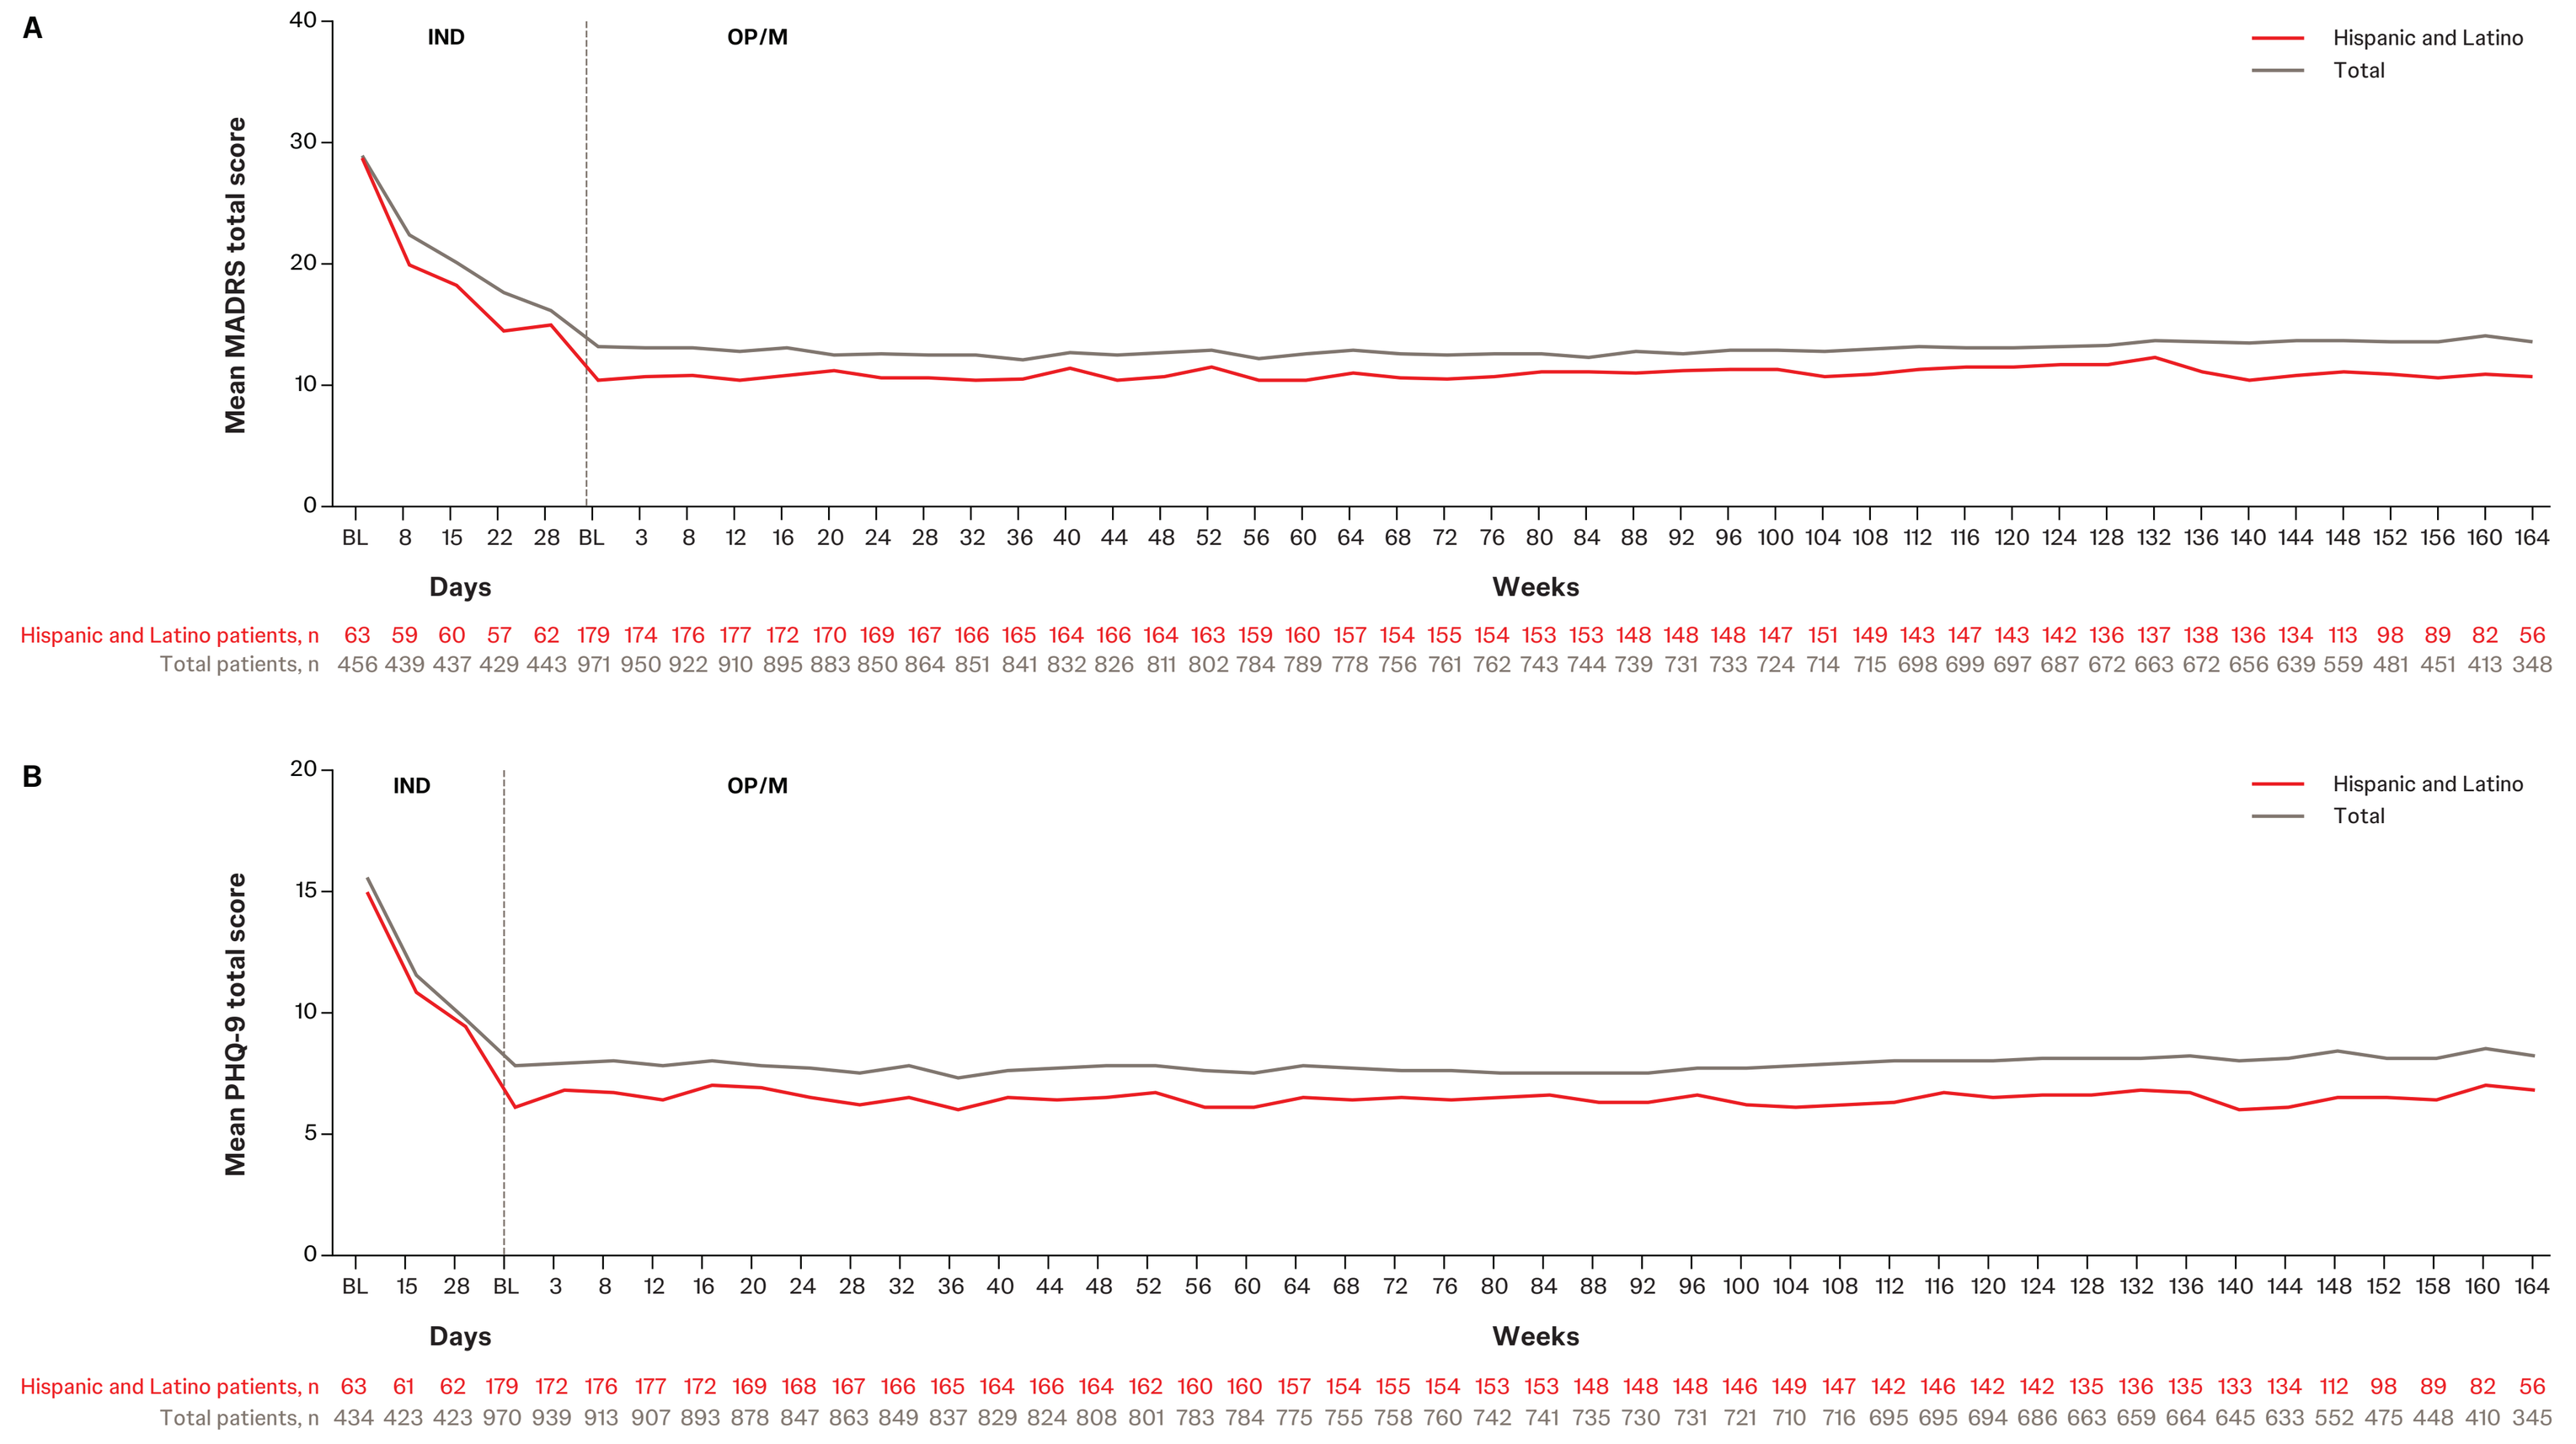
A
IND
OP/M
Hispanic and Latino
Total
Mean MADRS total score
Days
Weeks
Hispanic and Latino patients, n
Total patients, n
B
IND
OP/M
Hispanic and Latino
Total
Mean PHQ-9 total score
Days
Weeks
Hispanic and Latino patients, n
Total patients, n

BL, baseline; IND, induction; MADRS, Montgomery-Åsberg Depression Rating Scale; OP/M, optimization/maintenance; PHQ-9, Patient Health Questionnaire-9.

FIGURE 4: Mean SDS scores during the IND and OP/M phases (observed cases)

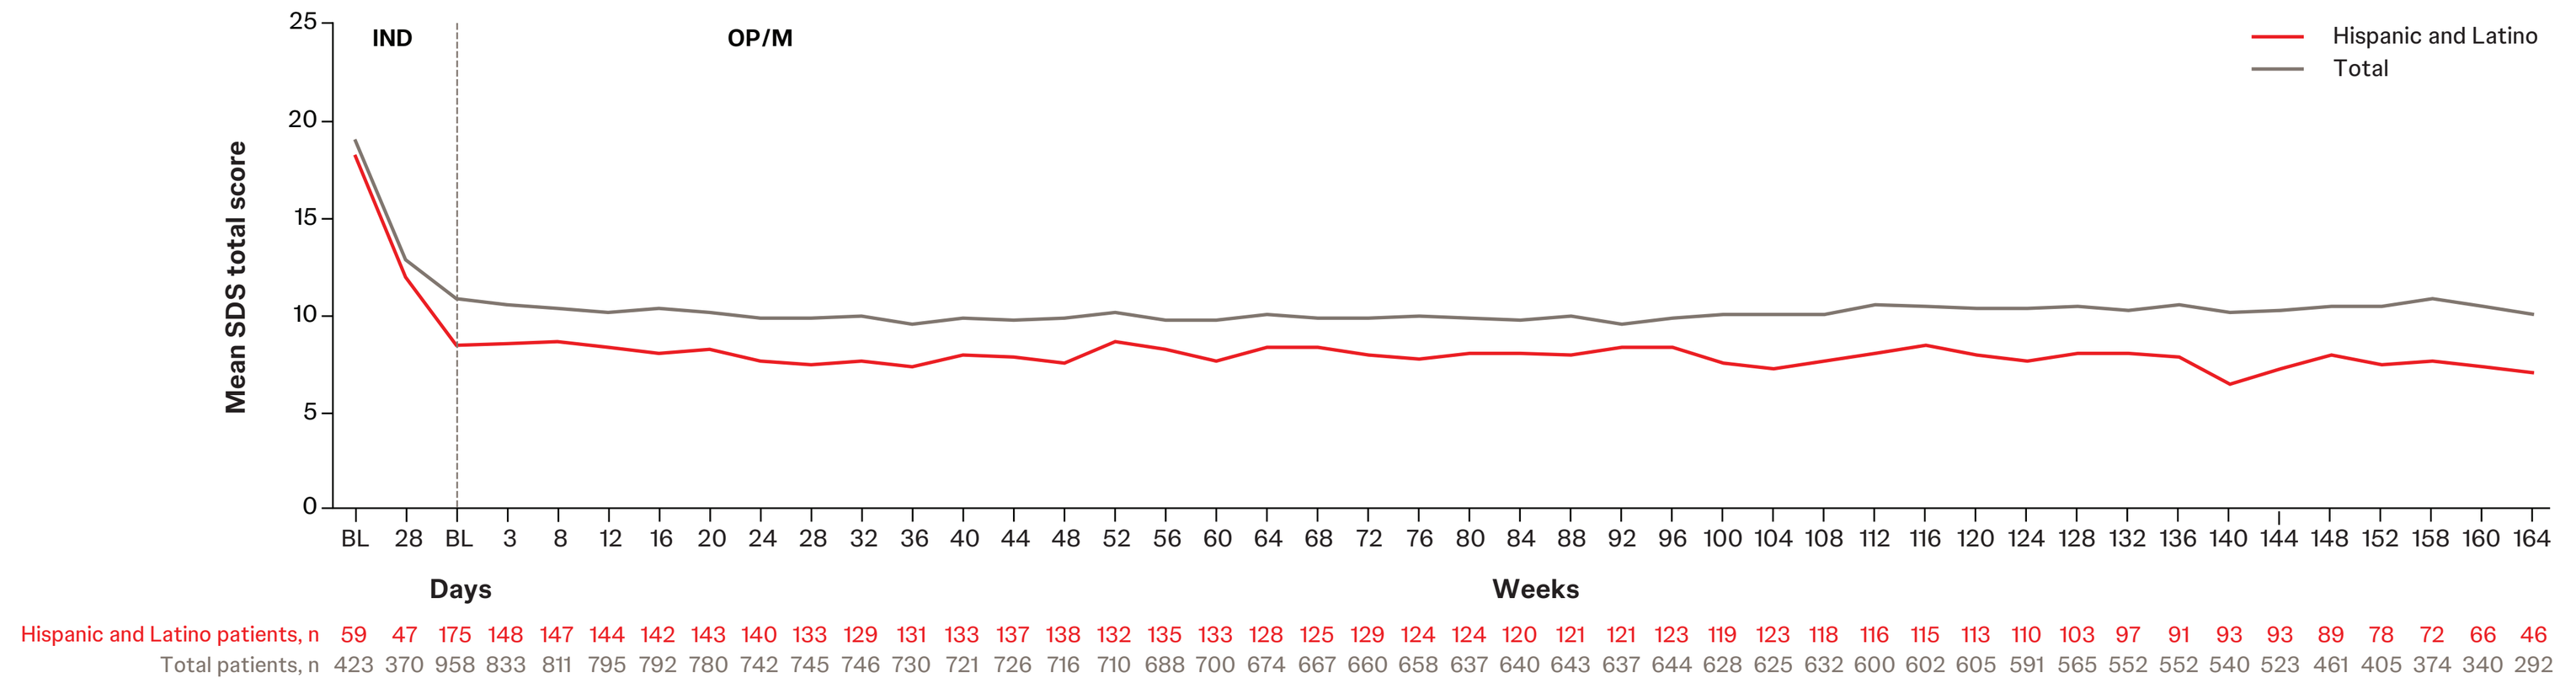
IND
OP/M
Hispanic and Latino
Total
Mean SDS total score
Days
Weeks
Hispanic and Latino patients, n
Total patients, n

BL, baseline; IND, induction; OP/M, optimization/maintenance; SDS, Sheehan Disability Scale.

Key Takeaways

- In SUSTAIN-3, Hispanic and Latino patients with treatment-resistant depression who were treated with esketamine nasal spray, in conjunction with an oral antidepressant, demonstrated improvements in depression symptom severity and functioning that were maintained throughout the study
- Long-term safety in Hispanic and Latino patients was consistent with the established safety and tolerability profiles of esketamine nasal spray

Limitations

- SUSTAIN-3 was an open-label study with no control group for comparison
- This was a subgroup analysis of Hispanic and Latino patients in SUSTAIN-3, which was not designed to specifically assess ESK treatment among this patient population
- As in the overall SUSTAIN-3 analysis, the generalizability of these findings may be limited by the exclusion of patients with significant psychiatric or medical comorbidities, the exclusion of patients with substance dependence, and potential bias related to which patients chose to continue treatment from the parent study

Conclusions

- The long-term safety of flexibly dosed esketamine nasal spray, in conjunction with an oral antidepressant, in Hispanic and Latino patients with treatment-resistant depression was consistent with the established safety profile of esketamine nasal spray and with safety results in the overall study population[4,6]
- Hispanic and Latino adults with treatment-resistant depression who continued their treatment in SUSTAIN-3 demonstrated improvements in depression symptom severity and functionality that were maintained throughout the study

Acknowledgements
The authors thank Lynn Brown, PhD, and Emily Spaulding, PhD (ApotheCom, Yardley, PA), for editorial and writing assistance, which was funded by Johnson & Johnson.

Disclosures
LH reports serving on advisory and steering committees for GH Research; consulting for AbbVie and Eli Lilly; and serving on the Janssen Neuroscience Advisory Panel on Mood Disorders and as a speaker for Janssen Neuroscience. IT, LM, and CC are employees of Johnson & Johnson and may hold stock or restricted stock units in the company.

Novel Pathways in Depression

Scan the QR code

The QR code is intended to provide scientific information for individual reference, and the information should not be altered or reproduced in any way.

This study was funded by Johnson & Johnson.

Presented at the American Psychiatric Association Annual Meeting; May 16-20, 2026; San Francisco, California, USA

References

1. US Department of Health and Human Services. *Key Substance Use and Mental Health Indicators in the United States: Results From the 2021 National Survey on Drug Use and Health*. US Department of Health and Human Services; 2022. HHS publication PEP22-07-01-005. Accessed February 2, 2026. https://www.samhsa.gov/data/report/2021-nsduh-annual-national-report 2. National Alliance on Mental Illness. Hispanic/Latinx. Accessed February 2, 2026. https://www.nami.org/your-journey/identity-and-cultural-dimensions/hispanic-latinx/ 3. Lubloy A. *BMC Health Serv Res*. 2014;14:469. 4. SPRAVATO® (esketamine) nasal spray, CIII. Prescribing information. Janssen Pharmaceuticals, Inc.; 2025. Accessed March 5, 2025. www.janssenlabels.com/package-insert/product-monograph/prescribing-information/SPRAVATO-pi.pdf 5. Zaki N et al. *Neuropsychopharmacology*. 2023;48(8):1225-1233. 6. Zaki N et al. *Int J Neuropsychopharmacol*. 2025;28(6):pyaf027.