251

Economic Burden of Patients with Recurrent and Metastatic Head and Neck Squamous Cell Carcinoma in the US

Jessica Geiger, MD[1], Safalta Khadka, PhD[2], Anne DePriest, PharmD[2], Yan Wang, ScD[3], Qian Shi, PhD[2], Cheryl Xiang, MBA[4], Dexter Waters, MS, MPH[2]

[1] Cleveland Clinic, Cleveland, OH, USA; [2] Johnson & Johnson Innovative Medicine, Titusville, NJ, USA; [3] Analysis Group, Inc., Los Angeles, CA, USA; [4] Analysis Group, Inc., Boston, MA, USA

Key takeaway

Among patients with R/M HNSCC, treatment patterns became more heterogeneous in later lines, while HRU and supportive care burden increased across successive LOTs, underscoring substantial unmet needs among patients progressing to later-line therapy

Conclusions

In real-world practice, PD-1 inhibitor-containing regimens were commonly used as 1L treatment for R/M HNSCC, with greater use of chemotherapy alone and EGFR-containing regimens in later LOTs

HRU and palliative care use increased across successive LOTs, with HNSCC-related HRU accounting for more than half of overall utilization and increasing in later lines, reflecting greater healthcare and supportive care needs among patients receiving later-line treatment

These findings highlight the continued need for more effective treatment options that may delay disease progression and reduce the disease and healthcare burden among patients who progress to later lines of therapy

Please scan QR code

Poster

The QR code is intended to provide scientific information for individual reference, and the information should not be altered or reproduced in any way. Additional information is provided here: https://www.congresshub.com/

Disclosures
This study was funded by Johnson & Johnson Innovative Medicine, Titusville, NJ, USA.

Background

- Recurrent/metastatic head and neck squamous cell carcinoma (R/M HNSCC) is associated with poor prognosis, substantial disease burden, and impaired quality of life[1]
- The systemic treatment landscape for R/M HNSCC has evolved from predominantly platinum-based chemotherapy to immune checkpoint inhibitors-based therapies; in June 2019, pembrolizumab in combination with chemotherapy became established first-line (1L) treatment options[2]
- Despite these advances, most patients ultimately progress, and evidence describing disease burden across lines of therapy remains limited, particularly in later lines
- Characterizing treatment patterns and disease burden across lines of therapy using real-world data can help identify unmet needs of R/M HNSCC and inform future treatment strategies

Objective

- To describe real-world treatment patterns and healthcare resource utilization (HRU) across 1L, second line (2L), and 3L among US adults with R/M HNSCC

Methods

Data source

- This was a retrospective observational study using data from IQVIA PharMetrics® Plus Closed Health Plan Claims database between January 1, 2016 and July 15, 2025

Study population

- Adults with ≥2 HNSCC diagnoses were identified. Among them, patients with recurrent HNSCC were further identified if they had a ≥120-day treatment-free interval, and those with metastatic HNSCC were identified based on distant organ involvement at a known site
 - Patients meeting criteria for both recurrent and metastatic disease were randomly assigned to one cohort to avoid double counting
- Patients were required to have initiated 1L treatment on or after July 1, 2019 following Food and Drug Administration (FDA) approval of pembrolizumab plus chemotherapy as 1L and have ≥3 months of follow-up, unless death occurred earlier
- Eligible patients with R/M HNSCC were classified into 1L, 2L, and 3L cohorts using a claims-based line of therapy (LOT) algorithm

Statistical analysis

- Patient characteristics during the 12-month baseline period before the 1L, 2L, and 3L initiation were described using descriptive statistics
- Treatment sequences from 1L to 3L were visualized using a Sankey diagram
- HRU on a per-person-per-year (PPPY) basis was evaluated during the 12-month baseline period before 1L initiation and during each LOT, from LOT initiation (index date) to the end of follow-up (earliest of end of continuous enrollment, one day before the next LOT for patients initiating a subsequent line, date of metastatic disease diagnosis for patients with recurrent disease, or death)
- A subgroup analysis of patients in the 3L cohort who received chemotherapy and pembrolizumab in 1L or 2L were conducted

Results

Figure 1. Sample selection flowchart of patients with R/M HNSCC receiving 1L-3L therapy[1]

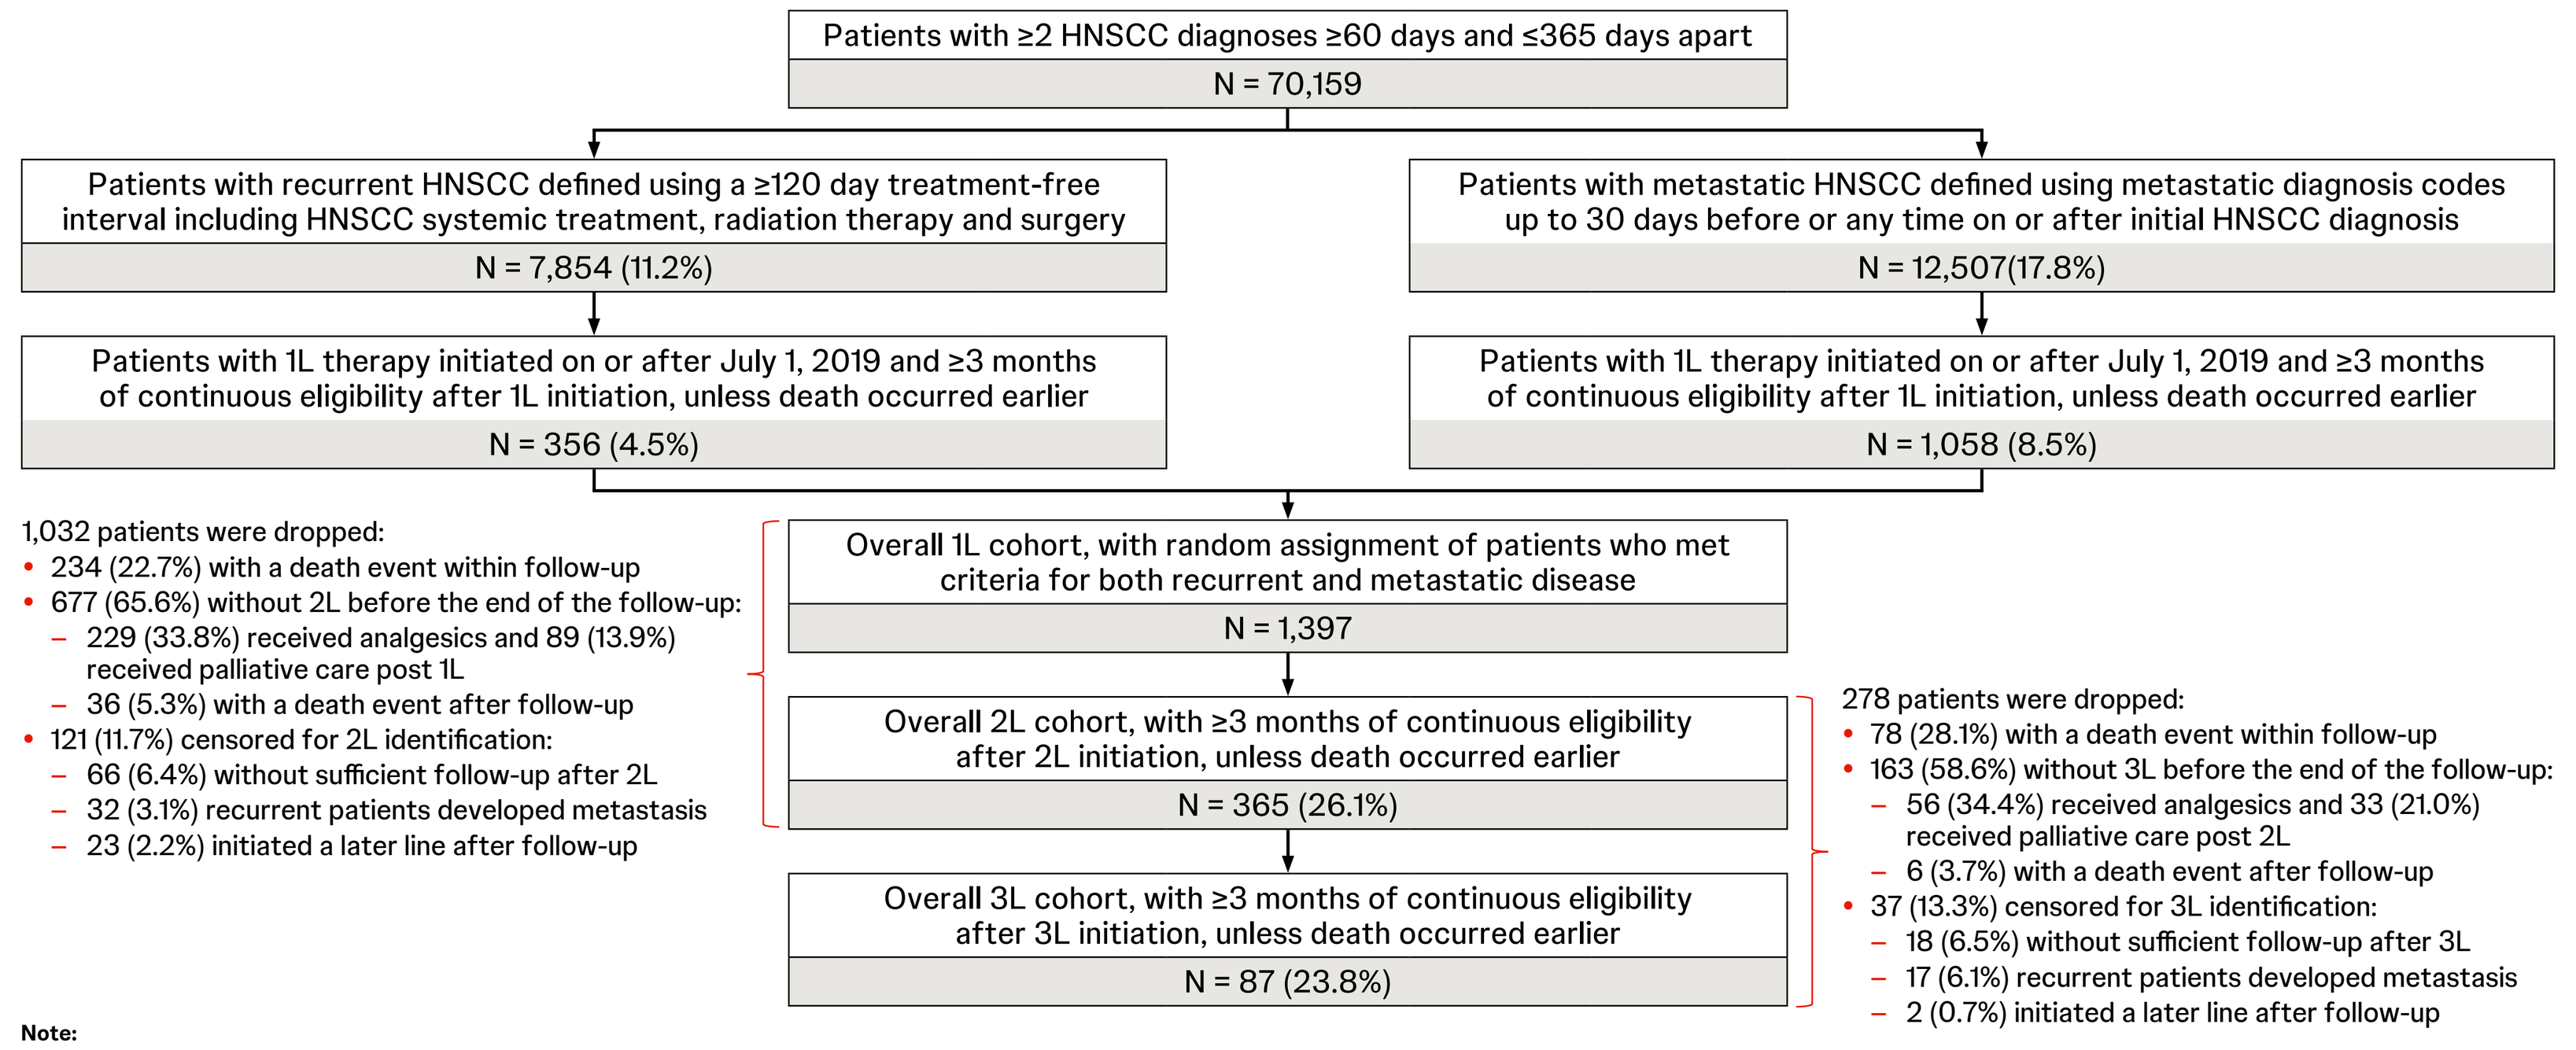
Patients with ≥2 HNSCC diagnoses ≥60 days and ≤365 days apart
N = 70,159
Patients with recurrent HNSCC defined using a ≥120 day treatment-free interval including HNSCC systemic treatment, radiation therapy and surgery
N = 7,854 (11.2%)
Patients with metastatic HNSCC defined using metastatic diagnosis codes up to 30 days before or any time on or after initial HNSCC diagnosis
N = 12,507(17.8%)
Patients with 1L therapy initiated on or after July 1, 2019 and ≥3 months of continuous eligibility after 1L initiation, unless death occurred earlier
N = 356 (4.5%)
Patients with 1L therapy initiated on or after July 1, 2019 and ≥3 months of continuous eligibility after 1L initiation, unless death occurred earlier
N = 1,058 (8.5%)
Overall 1L cohort, with random assignment of patients who met criteria for both recurrent and metastatic disease
N = 1,397
Overall 2L cohort, with ≥3 months of continuous eligibility after 2L initiation, unless death occurred earlier
N = 365 (26.1%)
Overall 3L cohort, with ≥3 months of continuous eligibility after 3L initiation, unless death occurred earlier
N = 87 (23.8%)
1,032 patients were dropped:
• 234 (22.7%) with a death event within follow-up
• 677 (65.6%) without 2L before the end of the follow-up:
– 229 (33.8%) received analgesics and 89 (13.9%) received palliative care post 1L
– 36 (5.3%) with a death event after follow-up
• 121 (11.7%) censored for 2L identification:
– 66 (6.4%) without sufficient follow-up after 2L
– 32 (3.1%) recurrent patients developed metastasis
– 23 (2.2%) initiated a later line after follow-up
278 patients were dropped:
• 78 (28.1%) with a death event within follow-up
• 163 (58.6%) without 3L before the end of the follow-up:
– 56 (34.4%) received analgesics and 33 (21.0%) received palliative care post 2L
– 6 (3.7%) with a death event after follow-up
• 37 (13.3%) censored for 3L identification:
– 18 (6.5%) without sufficient follow-up after 3L
– 17 (6.1%) recurrent patients developed metastasis
– 2 (0.7%) initiated a later line after follow-up

Note:
1. Systemic treatment for HNSCC include chemotherapies (cisplatin, carboplatin, 5-FU, paclitaxel, docetaxel, methotrexate, capecitabine, gemcitabine, etoposide, cyclophosphamide, doxorubicin, vincristine, hydroxyurea, pemetrexed, vinorelbine), EGFR-inhibitors (cetuximab, afatinib), PD-1 inhibitors (nivolumab, pembrolizumab), CTLA-4 inhibitors (ipilimumab), and targeted therapies (fam-trastuzumab deruxtecan-nxki, erdafitinib, dabrafenib, trametinib, entrectinib, larotrectinib, selpercatinib).

Baseline characteristics

- A total of 1,397, 365, and 87 patients were included in the 1L, 2L, and 3L cohort, respectively (**Figure 1**)
 - Among the 3L cohort, 66 patients (75.9%) were chemotherapy and pembrolizumab experienced in the 1L or 2L
- Across cohorts, patients had a mean age of 65.5 years at 1L, 65.0 years at 2L, and 63.4 years at 3L (**Table 1**)
- Oropharyngeal tumors were the most common tumor location at index, accounting for 48.5%, 54.5%, and 64.4% in the 1L, 2L, and 3L cohorts, respectively
- Over half of patients had lymph node involvement across cohorts, with increasing metastatic frequency in later lines

Table 1. Baseline characteristics

	Overall 1L Cohort N = 1,397	Overall 2L Cohort N = 365	Overall 3L Cohort N = 87	Chemotherapy and Pembrolizumab Experienced 3L Subgroup N = 66
Demographics				
Age at index (years), mean ± SD	65.5 ± 10.9	65.0 ± 10.7	63.4 ± 10.1	63.0 ± 10.1
Male, n (%)	1,073 (76.8%)	294 (80.5%)	72 (82.8%)	57 (86.4%)
Geographic region, n (%)				
Midwest	532 (38.1%)	130 (35.6%)	32 (36.8%)	25 (37.9%)
South	327 (23.4%)	90 (24.7%)	19 (21.8%)	18 (27.3%)
Northeast	316 (22.6%)	84 (23.0%)	20 (23.0%)	14 (21.2%)
West	195 (14.0%)	51 (14.0%)	13 (14.9%)	9 (13.6%)
Other	27 (1.9%)	10 (2.7%)	3 (3.4%)	0 (0.0%)
Insurance, n (%)				
Medicare Advantage	615 (44.0%)	151 (41.4%)	29 (33.3%)	20 (30.3%)
Commercial	483 (34.6%)	149 (40.8%)	45 (51.7%)	39 (59.1%)
Medicaid	287 (20.5%)	63 (17.3%)	12 (13.8%)	6 (9.1%)
Self-insured	12 (0.9%)	2 (0.5%)	1 (1.1%)	1 (1.5%)
Clinical characteristics				
Age at index HNSCC diagnosis (years), mean ± SD	63.9 ± 10.8	62.7 ± 10.6	60.8 ± 10.2	60.5 ± 10.1
Age at index recurrent or metastatic HNSCC diagnosis (years), mean ± SD	65.4 ± 10.9	64.1 ± 10.7	62.1 ± 10.2	61.7 ± 10.1
Modified CCI, mean ± SD	1.5 ± 1.6	1.7 ± 1.7	1.8 ± 1.5	1.7 ± 1.5
HNSCC tumor location closest to index				
Oropharynx	615 (48.5%)	187 (54.5%)	56 (64.4%)	42 (63.6%)
Lip and oral cavity	346 (27.3%)	83 (24.2%)	21 (24.1%)	16 (24.2%)
Larynx	294 (23.2%)	70 (20.4%)	13 (14.9%)	11 (16.7%)
Hypopharynx	58 (4.6%)	15 (4.4%)	3 (3.4%)	3 (4.5%)
Other and ill-defined sites in the lip, oral cavity and pharynx	48 (3.8%)	10 (2.9%)	2 (2.3%)	1 (1.5%)
Metastatic site				
Liver	164 (11.7%)	69 (18.9%)	29 (33.3%)	21 (31.8%)
Lung (lung, mediastinum, pleura)	461 (33.0%)	166 (45.5%)	60 (69.0%)	43 (65.2%)
Lymph node	717 (51.3%)	205 (56.2%)	46 (52.9%)	36 (54.5%)
Bone	274 (19.6%)	96 (26.3%)	28 (32.2%)	21 (31.8%)
Brain	49 (3.5%)	25 (6.8%)	7 (8.0%)	5 (7.6%)
Skin	27 (1.9%)	9 (2.5%)	5 (5.7%)	4 (6.1%)
Other	443 (31.7%)	164 (44.9%)	50 (57.5%)	37 (56.1%)
Prior treatment before index				
Any prior chemotherapy	555 (39.7%)	274 (75.1%)	81 (93.1%)	66 (100.0%)
Any prior PD-1 inhibitors	149 (10.7%)	263 (72.1%)	76 (87.4%)	66 (100.0%)

Note:
1. For metastatic sites, most of the "Other" category consists of "Other specified sites": 284 (20.3%) in 1L, 106 (29.0%) in 2L, and 33 (37.9%) in 3L.

Treatment patterns from 1L to 3L

- PD-1 inhibitor-containing regimens were common in 1L, with PD-1 inhibitor monotherapy accounting for 36.4% of 1L regimens, followed by PD-1 inhibitor-containing combination therapy at 28.8% (**Figure 2**)
- Chemotherapy alone was observed frequently across all regimens, increasing in prevalence from 27.9% in 1L to 37.5% in 2L and 46.0% in 3L
- EGFR-containing therapies were used more frequently in later lines, increasing from 6.7% in 1L to 32.1% in 2L and 32.2% in 3L
- Among chemotherapy and pembrolizumab-experienced patients with 3L regimens (N=66), the most common 3L regimens were chemotherapy (40.9%), EGFR + chemotherapy (19.7%), PD-1 inhibitors (12.1%), and pembrolizumab + chemotherapy (10.6%)

Figure 2. Treatment sequences from 1L to 3L

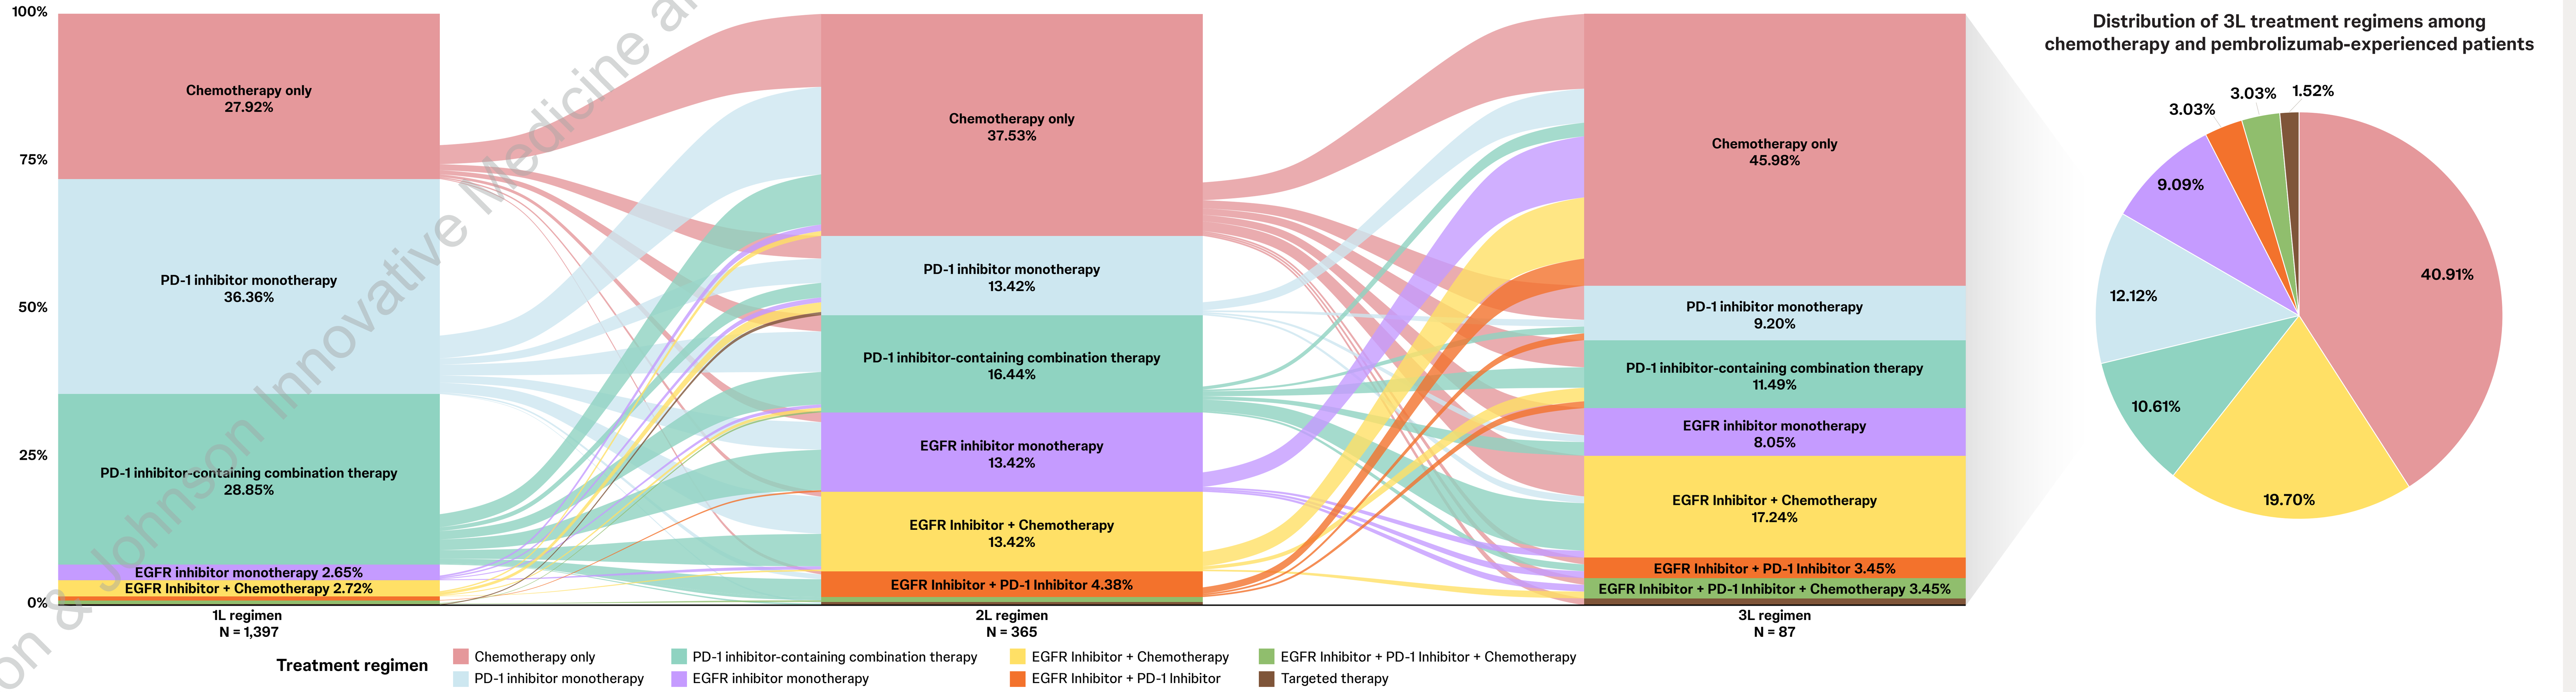
100%
75%
50%
25%
0%
Chemotherapy only 27.92%
PD-1 inhibitor monotherapy 36.36%
PD-1 inhibitor-containing combination therapy 28.85%
EGFR inhibitor monotherapy 2.65%
EGFR Inhibitor + Chemotherapy 2.72%
1L regimen N = 1,397
Chemotherapy only 37.53%
PD-1 inhibitor monotherapy 13.42%
PD-1 inhibitor-containing combination therapy 16.44%
EGFR inhibitor monotherapy 13.42%
EGFR Inhibitor + Chemotherapy 13.42%
EGFR Inhibitor + PD-1 Inhibitor 4.38%
2L regimen N = 365
Chemotherapy only 45.98%
PD-1 inhibitor monotherapy 9.20%
PD-1 inhibitor-containing combination therapy 11.49%
EGFR inhibitor monotherapy 8.05%
EGFR Inhibitor + Chemotherapy 17.24%
EGFR Inhibitor + PD-1 Inhibitor 3.45%
EGFR Inhibitor + PD-1 Inhibitor + Chemotherapy 3.45%
3L regimen N = 87
Distribution of 3L treatment regimens among chemotherapy and pembrolizumab-experienced patients
40.91%
19.70%
10.61%
12.12%
9.09%
3.03%
3.03%
1.52%
Treatment regimen
Chemotherapy only
PD-1 inhibitor monotherapy
PD-1 inhibitor-containing combination therapy
EGFR inhibitor monotherapy
EGFR Inhibitor + Chemotherapy
EGFR Inhibitor + PD-1 Inhibitor
EGFR Inhibitor + PD-1 Inhibitor + Chemotherapy
Targeted therapy

Note:
1. In 1L, the majority of PD-1 inhibitor-containing combination therapy consisted of PD-1 inhibitor + chemotherapy (96.28%).

HRU

- All-cause HRU burden increased in later LOTs (**Figure 3a**):
 - Mean inpatient (IP) admissions rising from 0.6 at baseline to 1.8, 1.9, and 1.8 PPPY in 1L, 2L, and 3L, respectively
 - Outpatient (OP) visits and home care were the primary drivers of HRU, with all-cause rates increasing by LOT (OP: 39.8, 49.3, 53.1, and 57.1 PPPY; home care: 11.0, 20.4, 23.9, and 25.6 PPPY at baseline, 1L, 2L, and 3L, respectively)
- More than 50% of HRU was attributable to HNSCC-related care, with the proportion increasing in later LOTs (**Figure 3b**)
- Supportive care also increased, with mean palliative care visits and PEG-tube-related visits rising from baseline to later lines (palliative care: 0.4, 2.2, 3.4, 4.9; PEG tube: 1.2, 2.7, 2.6, 3.2) (**Figure 3c-d**)
- The HNSCC-related burden during 3L was numerically higher in the chemotherapy and pembrolizumab-experienced patients (N=66) compared to the overall 3L cohort

Figure 3a. All-cause HRU

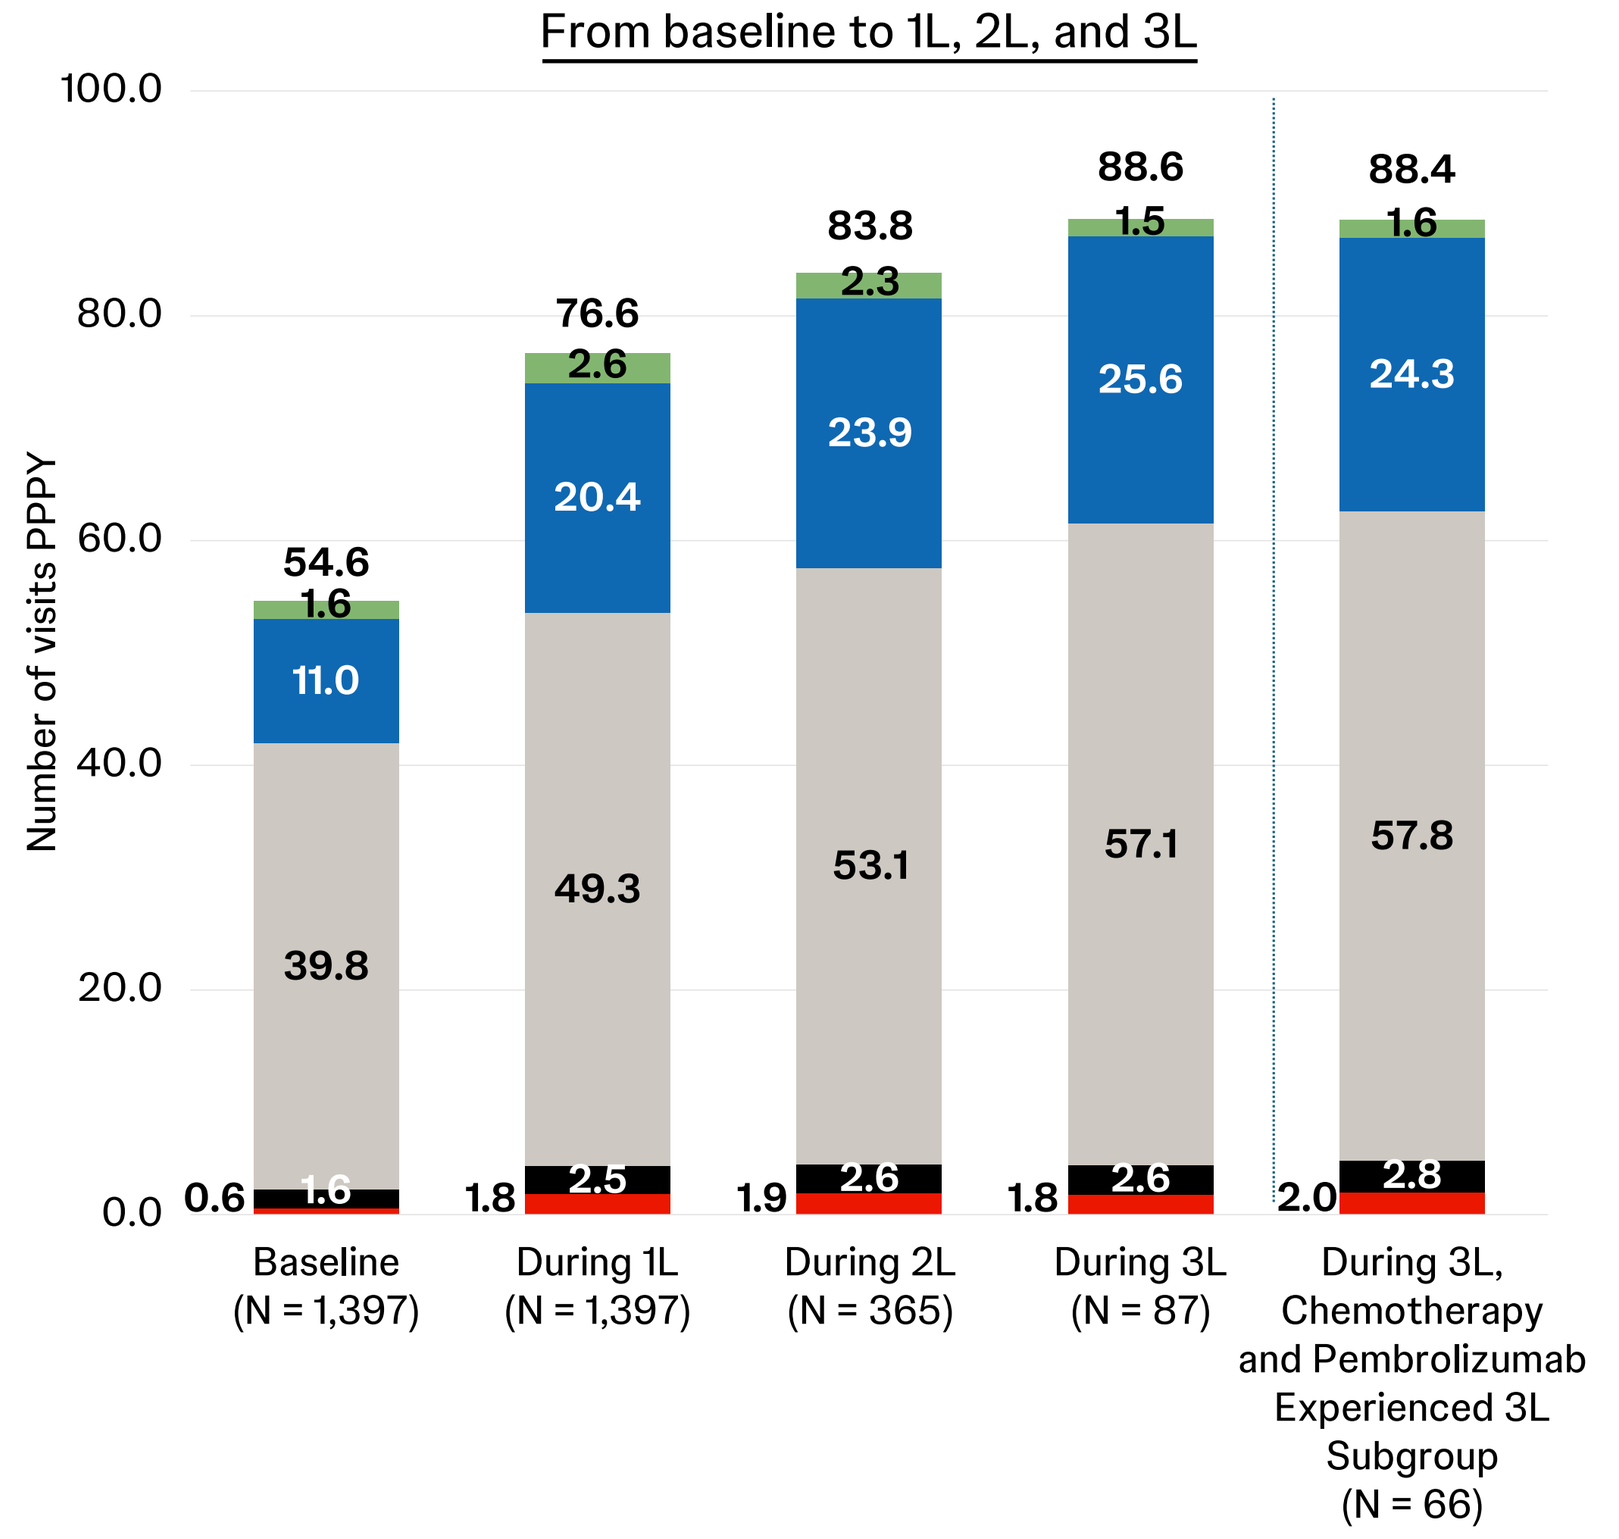
From baseline to 1L, 2L, and 3L
Number of visits PPPY
Baseline (N = 1,397): 54.6; 0.6, 1.6, 39.8, 11.0, 1.6
During 1L (N = 1,397): 76.6; 1.8, 2.5, 49.3, 20.4, 2.6
During 2L (N = 365): 83.8; 1.9, 2.6, 53.1, 23.9, 2.3
During 3L (N = 87): 88.6; 1.8, 2.6, 57.1, 25.6, 1.5
During 3L, Chemotherapy and Pembrolizumab Experienced 3L Subgroup (N = 66): 88.4; 2.0, 2.8, 57.8, 24.3, 1.6

Figure 3b. HNSCC-related HRU

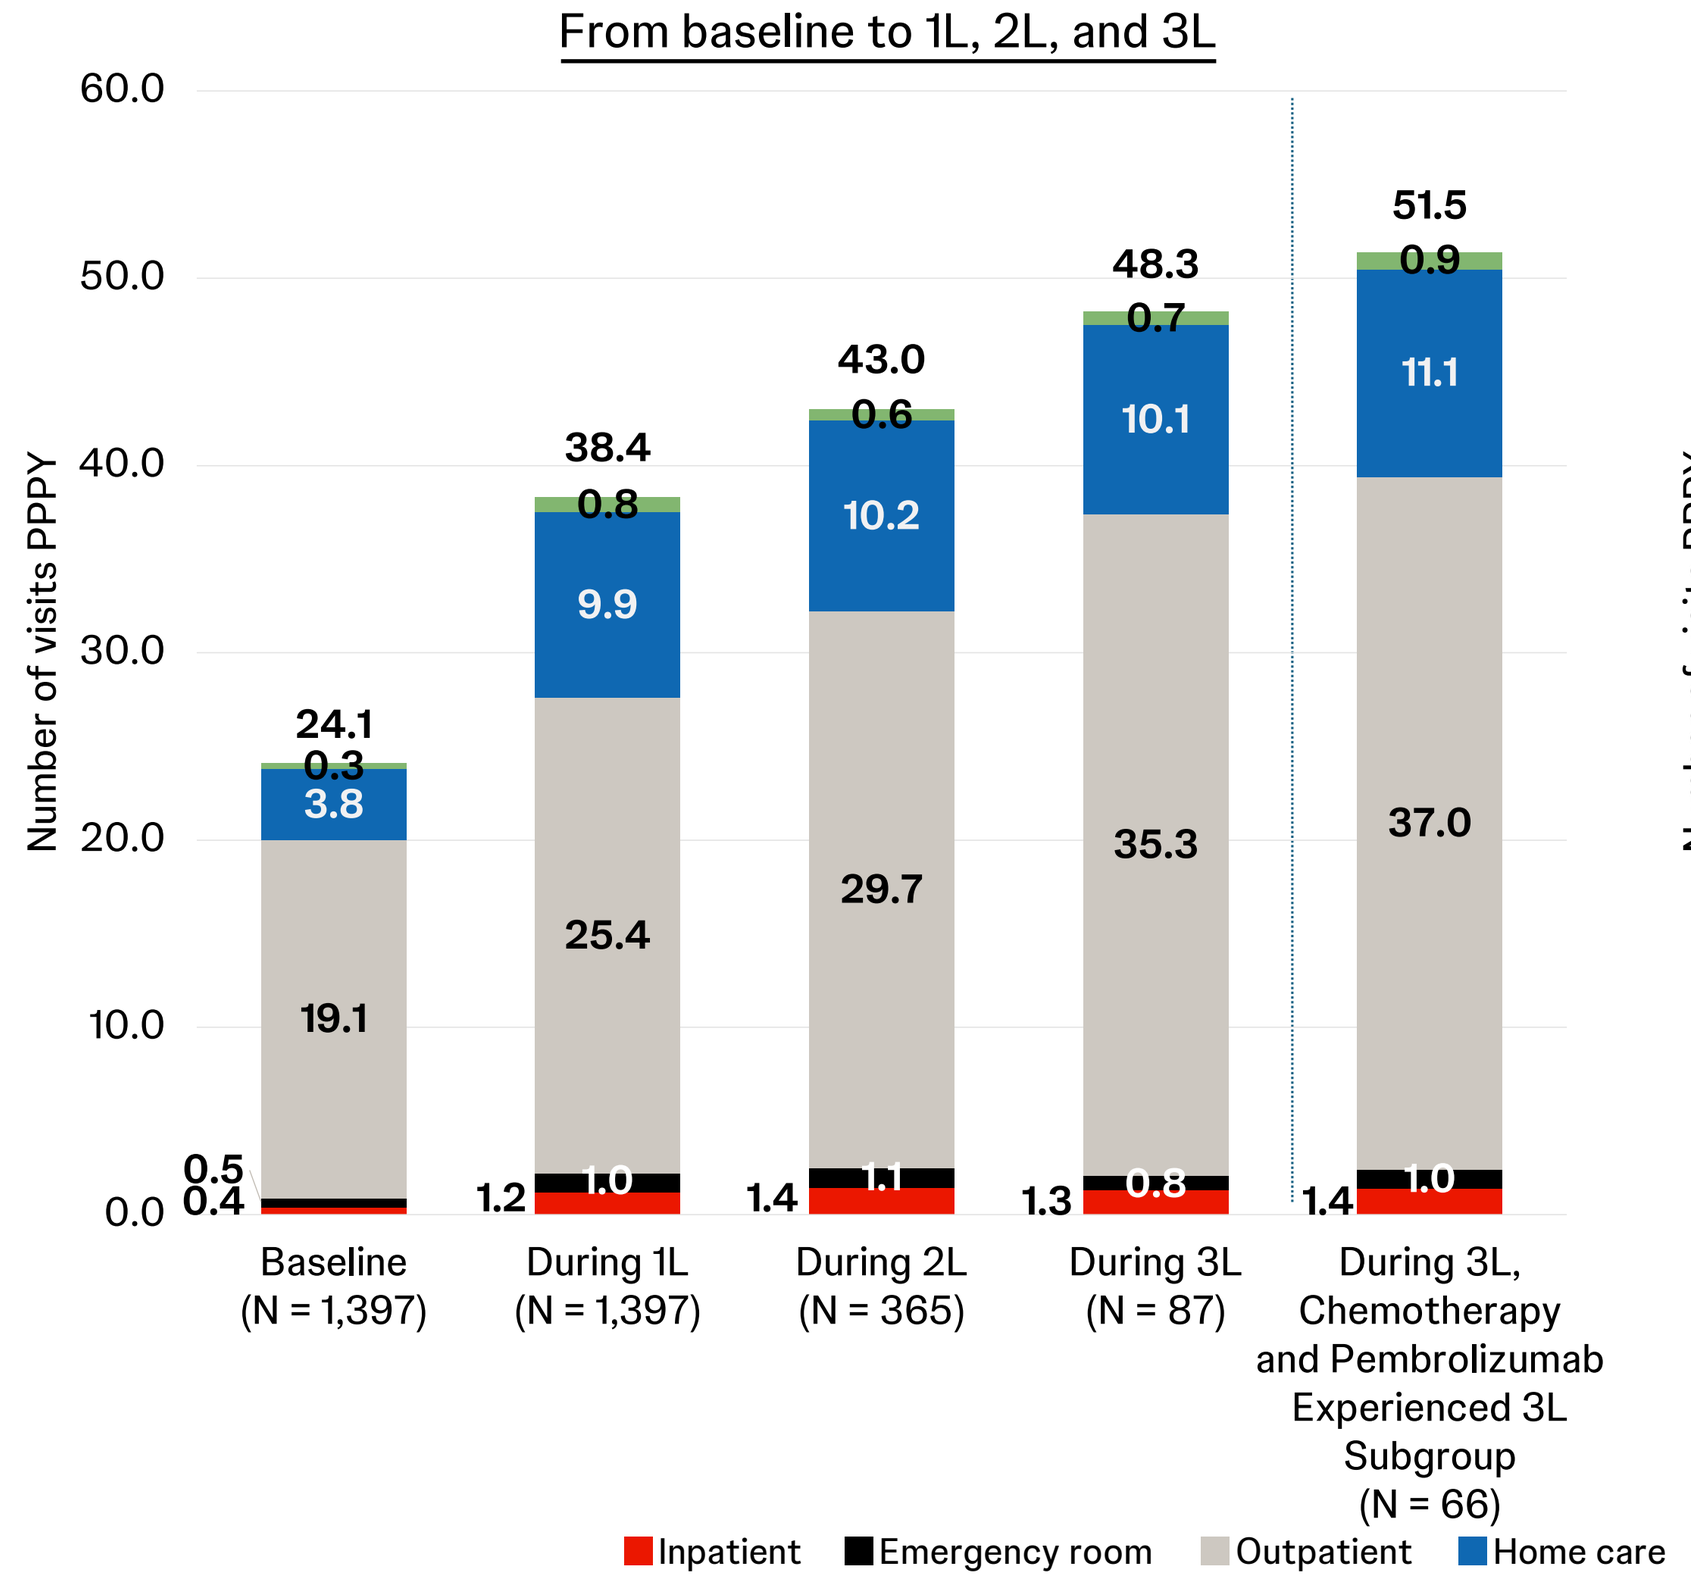
From baseline to 1L, 2L, and 3L
Number of visits PPPY
Baseline (N = 1,397): 24.1; 0.5, 0.4, 19.1, 3.8, 0.3
During 1L (N = 1,397): 38.4; 1.2, 1.0, 25.4, 9.9, 0.8
During 2L (N = 365): 43.0; 1.4, 1.1, 29.7, 10.2, 0.6
During 3L (N = 87): 48.3; 1.3, 0.8, 35.3, 10.1, 0.7
During 3L, Chemotherapy and Pembrolizumab Experienced 3L Subgroup (N = 66): 51.5; 1.4, 1.0, 37.0, 11.1, 0.9
Inpatient
Emergency room
Outpatient
Home care
Other (durable medical equipment, dental and vision care)

Figure 3c. Palliative care-related HRU

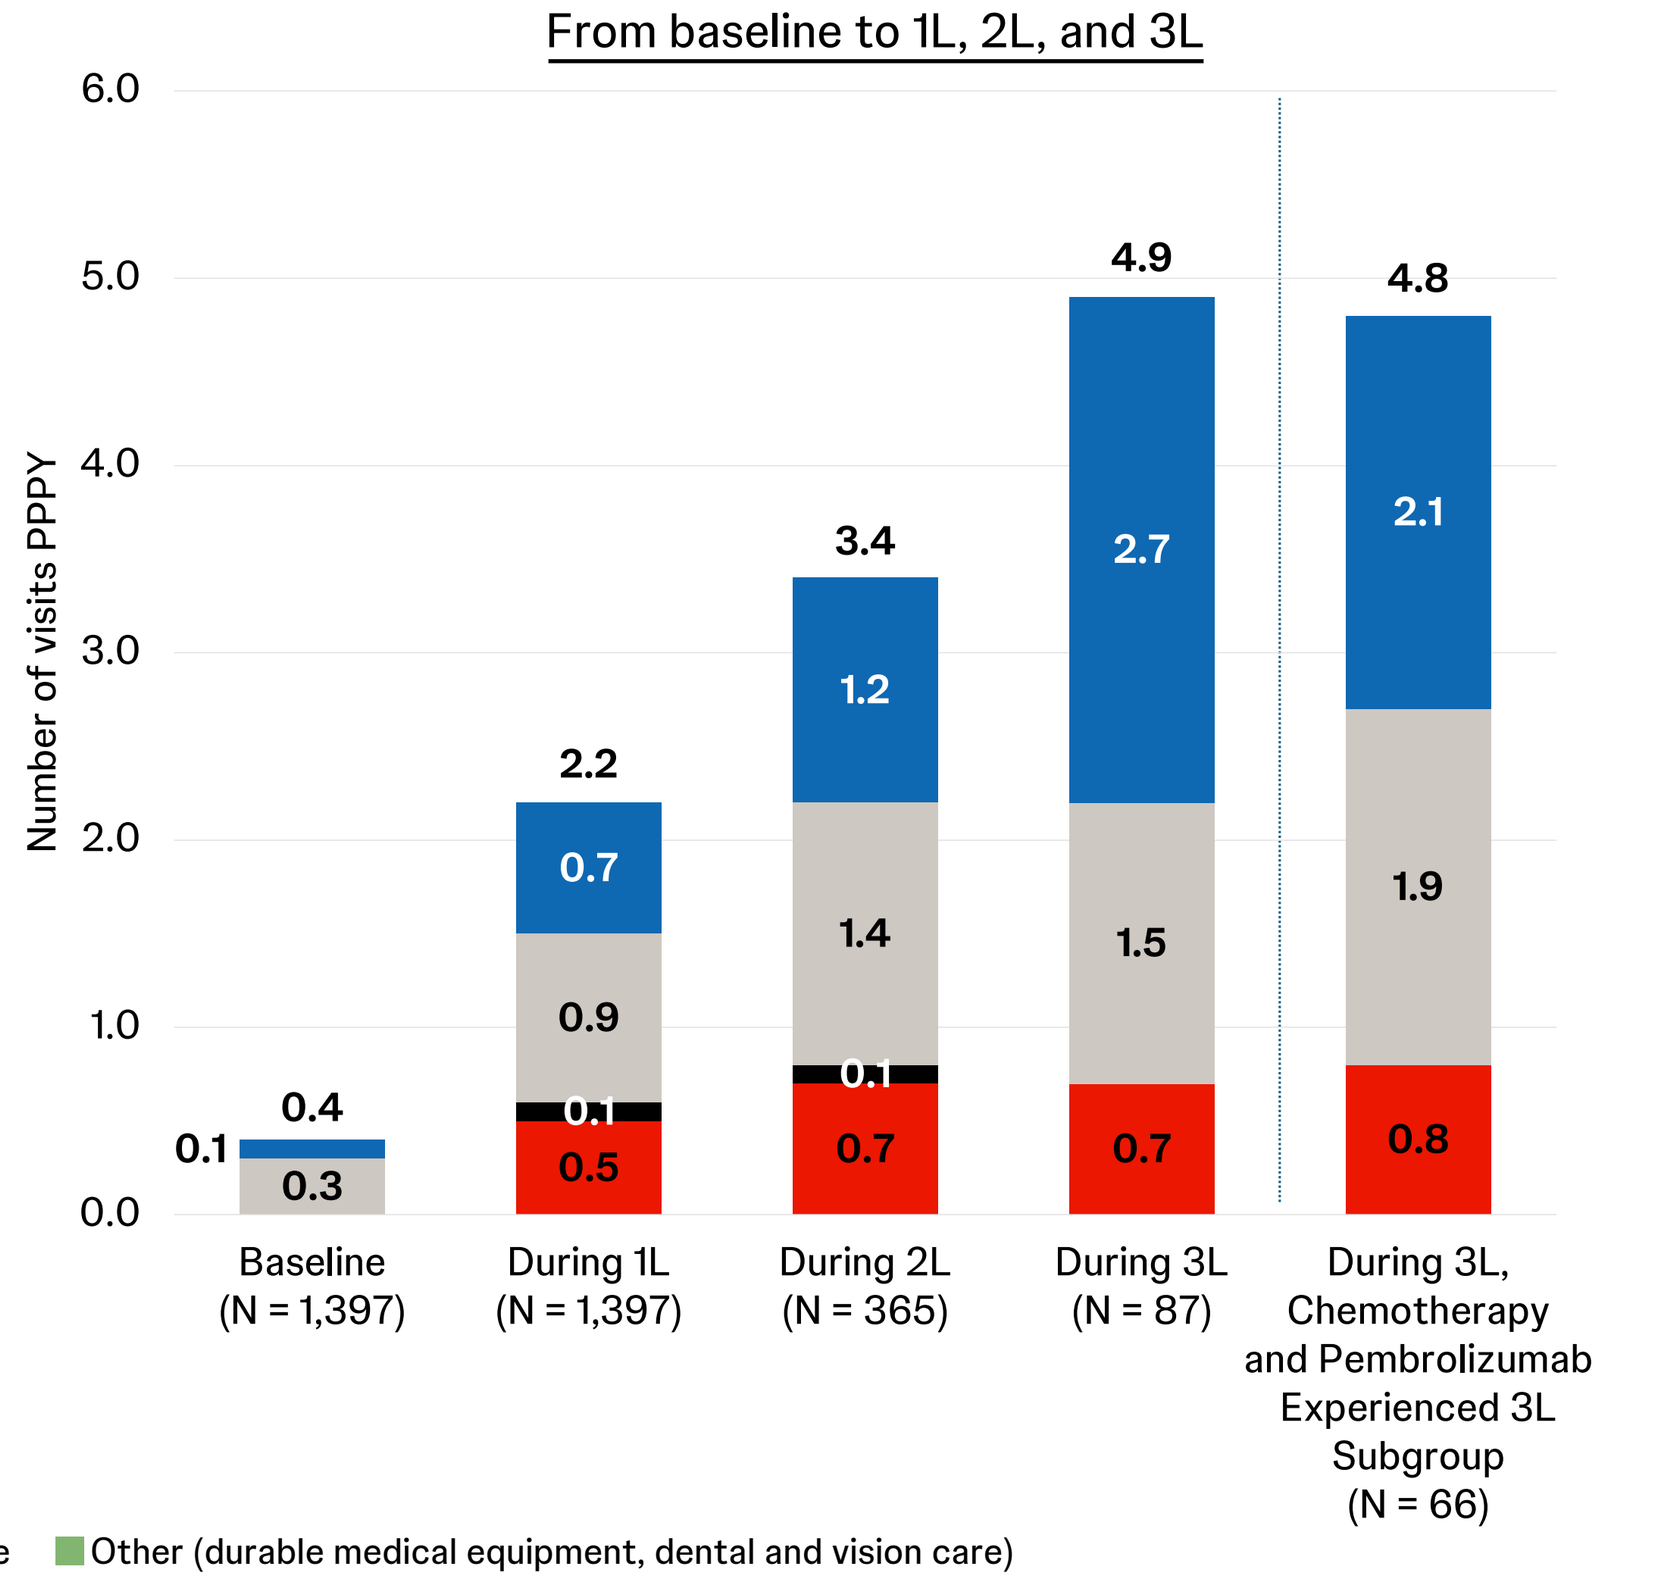
From baseline to 1L, 2L, and 3L
Number of visits PPPY
Baseline (N = 1,397): 0.4; 0.3, 0.1
During 1L (N = 1,397): 2.2; 0.5, 0.1, 0.9, 0.7
During 2L (N = 365): 3.4; 0.7, 0.1, 1.4, 1.2
During 3L (N = 87): 4.9; 0.7, 1.5, 2.7
During 3L, Chemotherapy and Pembrolizumab Experienced 3L Subgroup (N = 66): 4.8; 0.8, 1.9, 2.1

Figure 3d. PEG tube-related HRU

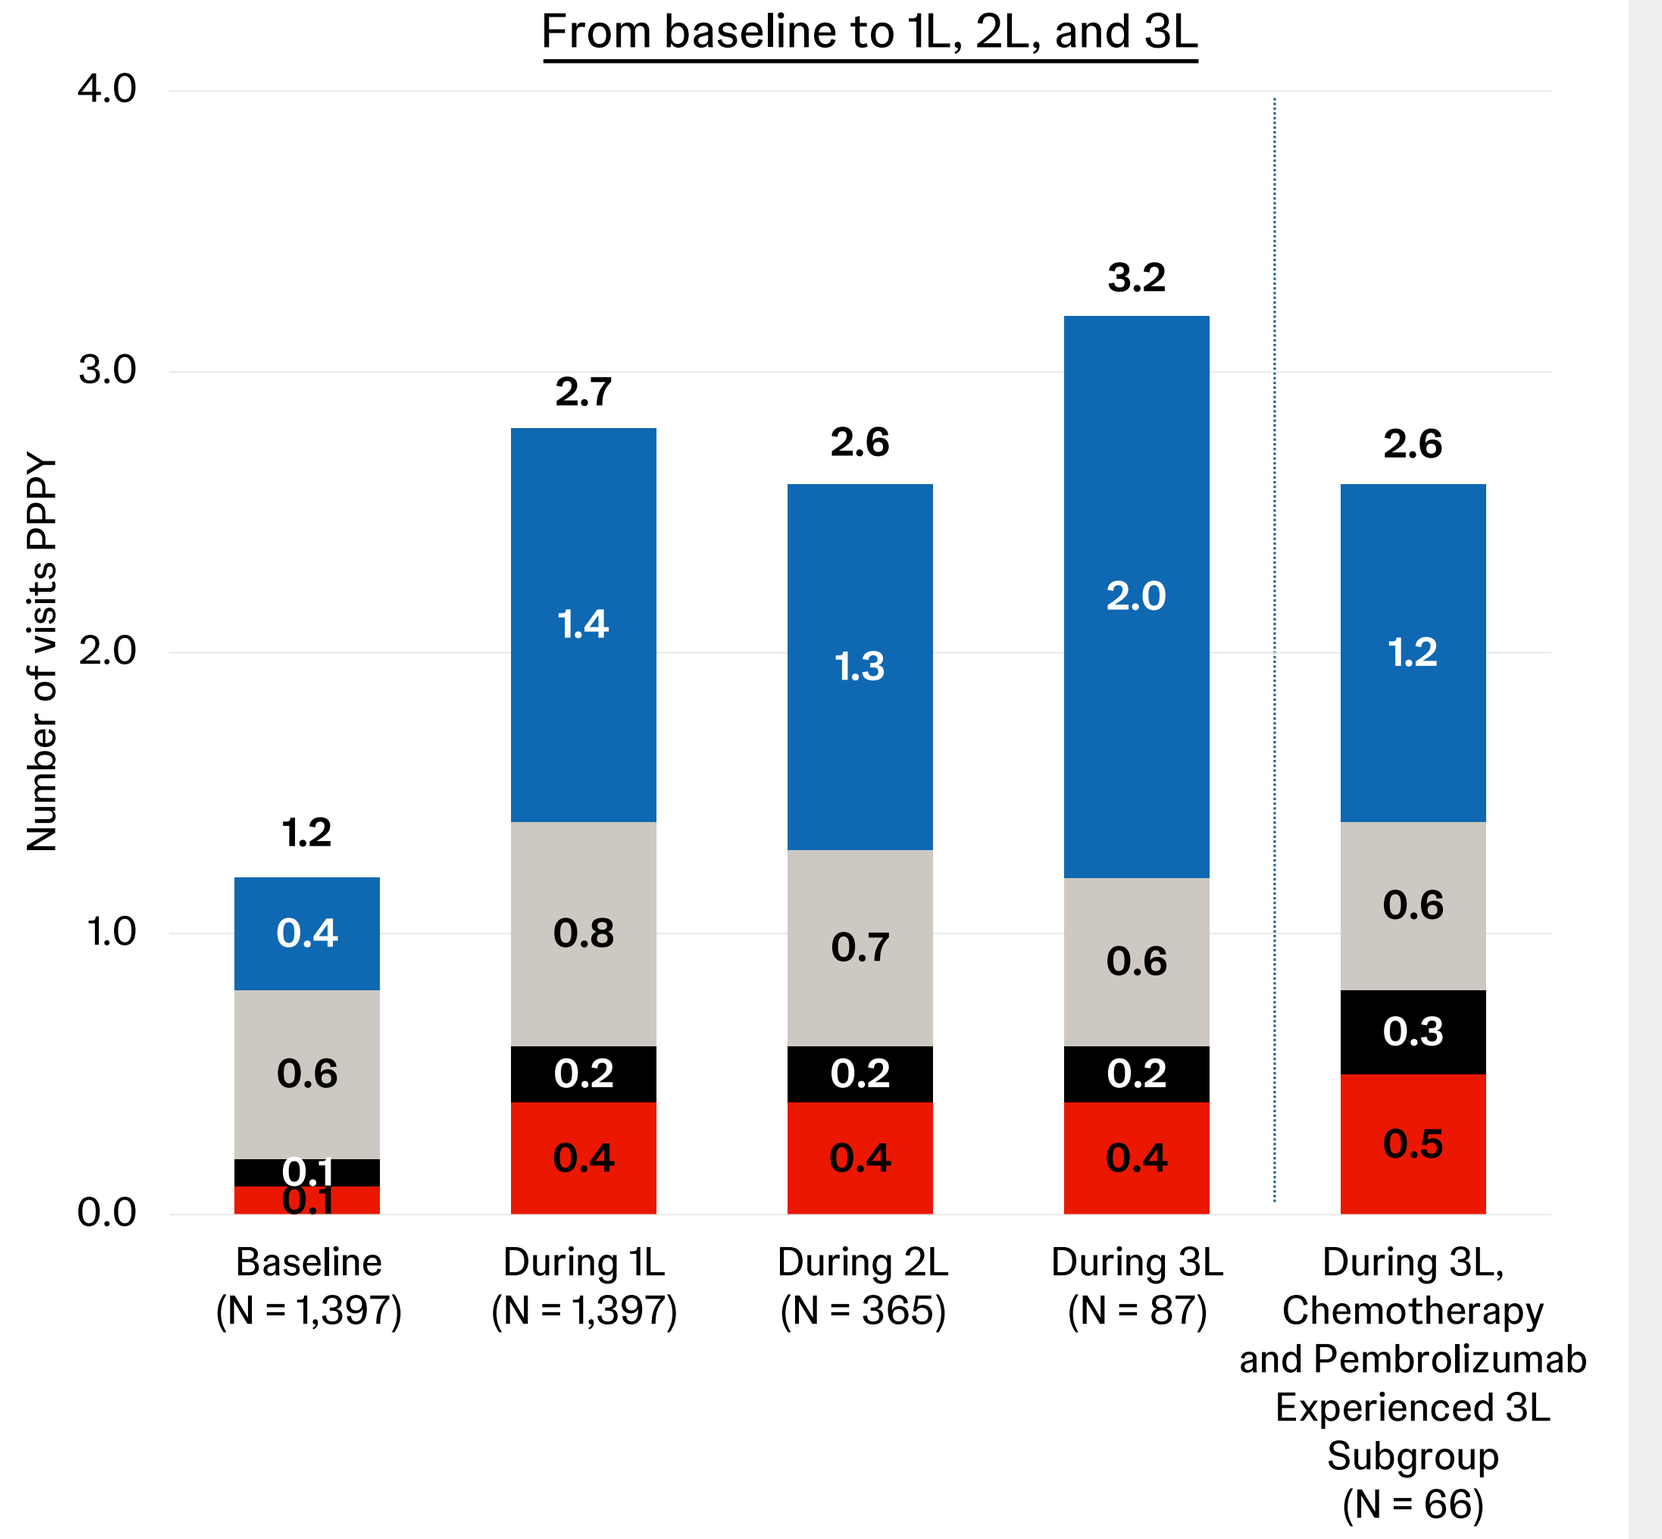
From baseline to 1L, 2L, and 3L
Number of visits PPPY
Baseline (N = 1,397): 1.2; 0.1, 0.1, 0.6, 0.4
During 1L (N = 1,397): 2.7; 0.4, 0.2, 0.8, 1.4
During 2L (N = 365): 2.6; 0.4, 0.2, 0.7, 1.3
During 3L (N = 87): 3.2; 0.4, 0.2, 0.6, 2.0
During 3L, Chemotherapy and Pembrolizumab Experienced 3L Subgroup (N = 66): 2.6; 0.5, 0.3, 0.6, 1.2

Notes:
1. The baseline period was the 12-month period before the index date. 2. The average 1L follow-up period was 1.0 years. 3. The average 2L follow-up period was 0.7 years. 4. The average 3L follow-up period was 0.6 years.

References
1. Degboe, Arnold, et al. *Journal of patient-reported outcomes* 2.1 (2018): 33.
2. Faraji, Farhoud, et al. *Translational Cancer Research* 12.5 (2023): 1353.

Presented at the Academy of Managed Care Pharmacy (AMCP) Nexus; October 26–29, 2026; San Diego, CA, USA

This material is distributed for scientific purposes by Johnson & Johnson Innovative Medicine and is not for promotional use.