338

Association of Hospitalization History Prior to Long-Acting Injectable Antipsychotic Initiation and Subsequent Hospitalizations and Costs in Young Dual-Eligibles With Schizophrenia: The CRITICAL PERIOD Study

Pengxiang Li[1], Zhi Geng[1], Carmela Benson[2], Jalpa A. Doshi[1,3]

[1]Division of General Internal Medicine, University of Pennsylvania, Philadelphia, PA; [2]Johnson & Johnson, Titusville, NJ; [3]Leonard Davis Institute of Health Economics, Philadelphia, PA

Background

- Schizophrenia (SCZ) is a chronic, severe psychiatric disorder associated with substantial clinical, humanistic, and economic burden[1]
- Suboptimal adherence to oral antipsychotics (APs) is common and is a major contributor to relapses, emergency department visits, and SCZ-related hospitalizations[2–4]
- Real-world evidence has demonstrated that long-acting injectable antipsychotics (LAIs) are associated with improved medication adherence and a reduced risk of SCZ-related hospitalizations compared with oral APs[5,6]
- However, LAI initiation is often delayed in young people with SCZ[7]

Objective

- The CRITICAL PERIOD study intended to examine whether schizophrenia (SCZ)-related hospitalization history prior to first LAI initiation is associated with subsequent hospitalizations and healthcare costs among young dual-eligible adults in the United States

Methods

Data source

- Data were taken from 2006 to 2021 national Medicare and Medicaid fee-for-service (FFS) claims available from the Centers for Medicare and Medicaid Services

Sample selection

- All beneficiaries having ≥1 claim with International Classification of Diseases (ICD)-9 code or ICD-10 code of SCZ in any position from the Medicaid or Medicare FFS claims, with the first observed claims occurring between July 1, 2009, and December 31, 2021
- Dually eligible for Medicare and Medicaid when they were first eligible for Medicare
- ≥1 antipsychotic fill between July 1, 2009, and December 31, 2021, with the first AP fill occurring on or after the date of the first observed SCZ diagnosis date in Medicare or Medicaid claims
- Continuous full Medicaid (FFS or health maintenance organization [HMO]) coverage or Medicare FFS Part D coverage from first SCZ diagnosis date until first AP fill date (ie, index date)
- First eligible for Medicare due to disability (excluding patients aging into Medicare since they likely had private insurance prior to turning 65 years old)
- Use of an LAI between July 1, 2009, and December 31, 2021; the first date of LAI use was the index date
- Time between the first observed SCZ diagnosis date and first LAI use date (index date) was 1 month or more
- Continuous coverage of at least 12 months in the post-index period (to allow a 12-month fixed follow-up period to measure outcomes)
- Age ≤25 years when first diagnosed with schizophrenia

Independent variable

- Patients were stratified by the number of SCZ-related hospitalizations in the pre-index period (0, 1, 2-5, or ≥6 hospitalizations)

Outcomes

- All-cause costs during the 12-month post-index period
- Number of hospitalizations during the 12-month post-index period
 - All-cause hospitalizations
 - Mental health–related hospitalizations
 - SCZ-related hospitalizations

Analysis

- Descriptive analyses were conducted on patient characteristics and outcomes in the overall sample and by independent variable
- Ordinary least squares (OLS) models were used to assess the association between pre-index hospitalization category and post-index utilization and costs, adjusting for demographic and clinical characteristics

Results

- The final sample consisted of 3,447 patients initiating an LAI (**Table 1**)
- Patients had a similar mean age of 22 years on SCZ diagnosis date across groups; they were primarily male (78%) and non-White (61.7%, **Table 2**)
 - Over half (51%) of patients had 0 SCZ-related hospitalizations in the pre-index period; the remaining patients had 1 (22%), 2-5 (22%), and ≥6 (5%) SCZ-related hospitalizations, respectively
 - The median time from first observed SCZ diagnosis to first observed LAI initiation was 331 days (162 for 0, 373 for 1, 719 for 2-5, and 1,251 for ≥6 SCZ-related hospitalizations)
- In descriptive analysis, greater pre-index hospitalization burden was associated with higher post-index all-cause (0.6-2.9), mental health–related (0.5-2.8), and SCZ-related (0.4-2.4) hospitalizations and higher all-cause costs ($31,527-$59,066) (**Table 3, Figure 1**)
- OLS regressions confirmed these associations (**Table 4**)
 - For instance, patients with ≥6 vs 0 pre-index hospitalizations had higher post-index all-cause (+1.89, 95%CI: 1.57, 2.20), mental health–related (+1.86, 95%CI: 1.57, 2.16), and SCZ-related (+1.71, 95%CI: 1.47, 1.96) hospitalizations and higher all-cause costs +$20,092.86, 95%CI: 14,197.47, 25,988.26)

Table 1: Sample attrition

Criteria	N
All beneficiaries having ≥1 claim with ICD-9 code or ICD-10 code of SCZ in any position from the Medicaid or Medicare FFS claims, with the first observed claims occurring between July 1, 2009, and December 31, 2021	1,338,215
Dually eligible for Medicare and Medicaid when they were first eligible for Medicare	340,524
≥1 antipsychotic fill between July 1, 2009, and December 31, 2021, with the first AP fill date occurring on or after the date of the first observed SCZ diagnosis date in Medicare or Medicaid claims	86,110
Continuous full Medicaid (FFS or HMO) coverage or Medicare FFS Part D coverage from first SCZ diagnosis date till first AP fill date (ie, index date)	40,412
First eligible for Medicare due to disability (excluding patients aging into Medicare since they likely had private insurance prior to turning 65 years old)	33,102
Use of an LAI between July 1, 2009, and December 31, 2021; the first date of LAI use was the index date	11,735
Time between the first observed SCZ diagnosis date and first LAI use date (index date) was 1 month or more	10,808
Continuous coverage of at least 12 months in the post-index period (to allow a 12-month fixed follow-up period to measure outcomes)	9,472
Age ≤25 years when first diagnosed with schizophrenia	3,447

AP, antipsychotic; FFS, fee for service; HMO, health maintenance organization; ICD, International Classification of Diseases; LAI, long-acting injectable; SCZ, schizophrenia.

Table 2: Patient characteristics

	Overall (N = 3447)	Number of schizophrenia-related hospitalizations during the pre-index period			
		0 (n = 1750)	1 (n = 764)	2 to 5 (n = 761)	≥6 (n = 172)
Age on SCZ diagnosis date, years					
Mean (SD)	22.3 (2.0)	22.4 (2.0)	22.2 (2.0)	22.2 (2.0)	22.2 (2.0)
Median (IQR)	22.0 (21.0, 24.0)	23.0 (21.0, 24.0)	22.0 (21.0, 24.0)	22.0 (21.0, 24.0)	22.0 (21.0, 24.0)
Male, n (%)	2672 (77.5)	1388 (79.3)	586 (76.7)	577 (75.8)	121 (70.3)
Race, n (%)					
White	1168 (33.9)	571 (32.6)	267 (34.9)	260 (34.2)	70 (40.7)
Black	1111 (32.2)	521 (29.8)	255 (33.4)	268 (35.2)	67 (39.0)
Hispanic	323 (9.4)	188 (10.7)	55 (7.2)	NR	NR
Other	693 (20.1)	386 (22.1)	151 (19.8)	135 (17.7)	21 (12.2)
Missing	152 (4.4)	84 (4.8)	36 (4.7)	NR	NR
Urban, n (%)	2921 (84.7)	1468 (83.9)	647 (84.7)	654 (85.9)	152 (88.4)
Days from first observed schizophrenia diagnosis date to index date					
Mean (SD)	592.0 (686.0)	370.3 (529.1)	586.2 (645.1)	918.8 (735.4)	1427.6 (823.9)
Median (IQR)	331.0 (92.0, 848.0)	162.0 (43.0, 461.0)	373.0 (127.0, 822.5)	719.0 (346.0, 1280.0)	1251.5 (759.5, 1996.5)
Without relapses in 1-month pre-index period	2417 (70.1)	1422 (81.3)	491 (64.3)	428 (56.2)	76 (44.2)
OAP agents used in pre-index period, n (%)					
Any OAP use	2460 (71.4)	983 (56.2)	603 (78.9)	706 (92.8)	168 (97.7)
Haloperidol	683 (19.8)	195 (11.1)	163 (21.3)	244 (32.1)	81 (47.1)
Quetiapine	674 (19.6)	218 (12.5)	143 (18.7)	229 (30.1)	84 (48.8)
Olanzapine	871 (25.3)	277 (15.8)	188 (24.6)	307 (40.3)	99 (57.6)
Risperidone	1188 (34.5)	411 (23.5)	280 (36.6)	394 (51.8)	103 (59.9)
Fluphenazine	157 (4.6)	39 (2.2)	36 (4.7)	60 (7.9)	22 (12.8)
Aripiprazole	756 (21.9)	282 (16.1)	187 (24.5)	214 (28.1)	73 (42.4)
Clozapine	146 (4.2)	40 (2.3)	31 (4.1)	52 (6.8)	23 (13.4)
Ziprasidone	299 (8.7)	101 (5.8)	56 (7.3)	100 (13.1)	42 (24.4)
Paliperidone	371 (10.8)	148 (8.5)	78 (10.2)	118 (15.5)	27 (15.7)
Lurasidone	288 (8.4)	93 (5.3)	57 (7.5)	106 (13.9)	32 (18.6)
Any other agent	30 (0.9)	18 (1.0)	NR	NR	NR
Evidence of mental health-related hospitalization in 30-day pre-index period, n (%)	680 (19.7)	61 (3.5)	209 (27.4)	316 (41.5)	94 (54.7)

IQR, interquartile range; NR, not reached; OAP, oral antipsychotic; SCZ, schizophrenia.

Table 3: Number of hospitalizations over 12 months following LAI Initiation

	Overall (N = 3447)	Number of schizophrenia-related hospitalizations during the pre-index period			
		0 (n = 1750)	1 (n = 764)	2 to 5 (n = 761)	≥6 (n = 172)
Number of all-cause hospitalizations, mean (SD)	0.9 (1.9)	0.6 (1.3)	0.9 (1.6)	1.4 (2.4)	2.9 (3.5)
Number of mental health–related hospitalizations, mean (SD)	0.9 (1.8)	0.5 (1.1)	0.8 (1.5)	1.3 (2.2)	2.8 (3.4)
Number of schizophrenia-related hospitalizations, mean (SD)	0.7 (1.5)	0.4 (0.9)	0.6 (1.1)	1.1 (1.9)	2.4 (2.9)

LAI, long-acting injectable.

Figure 1: Total healthcare all-cause costs in US dollars over 12 months following LAI Initiation

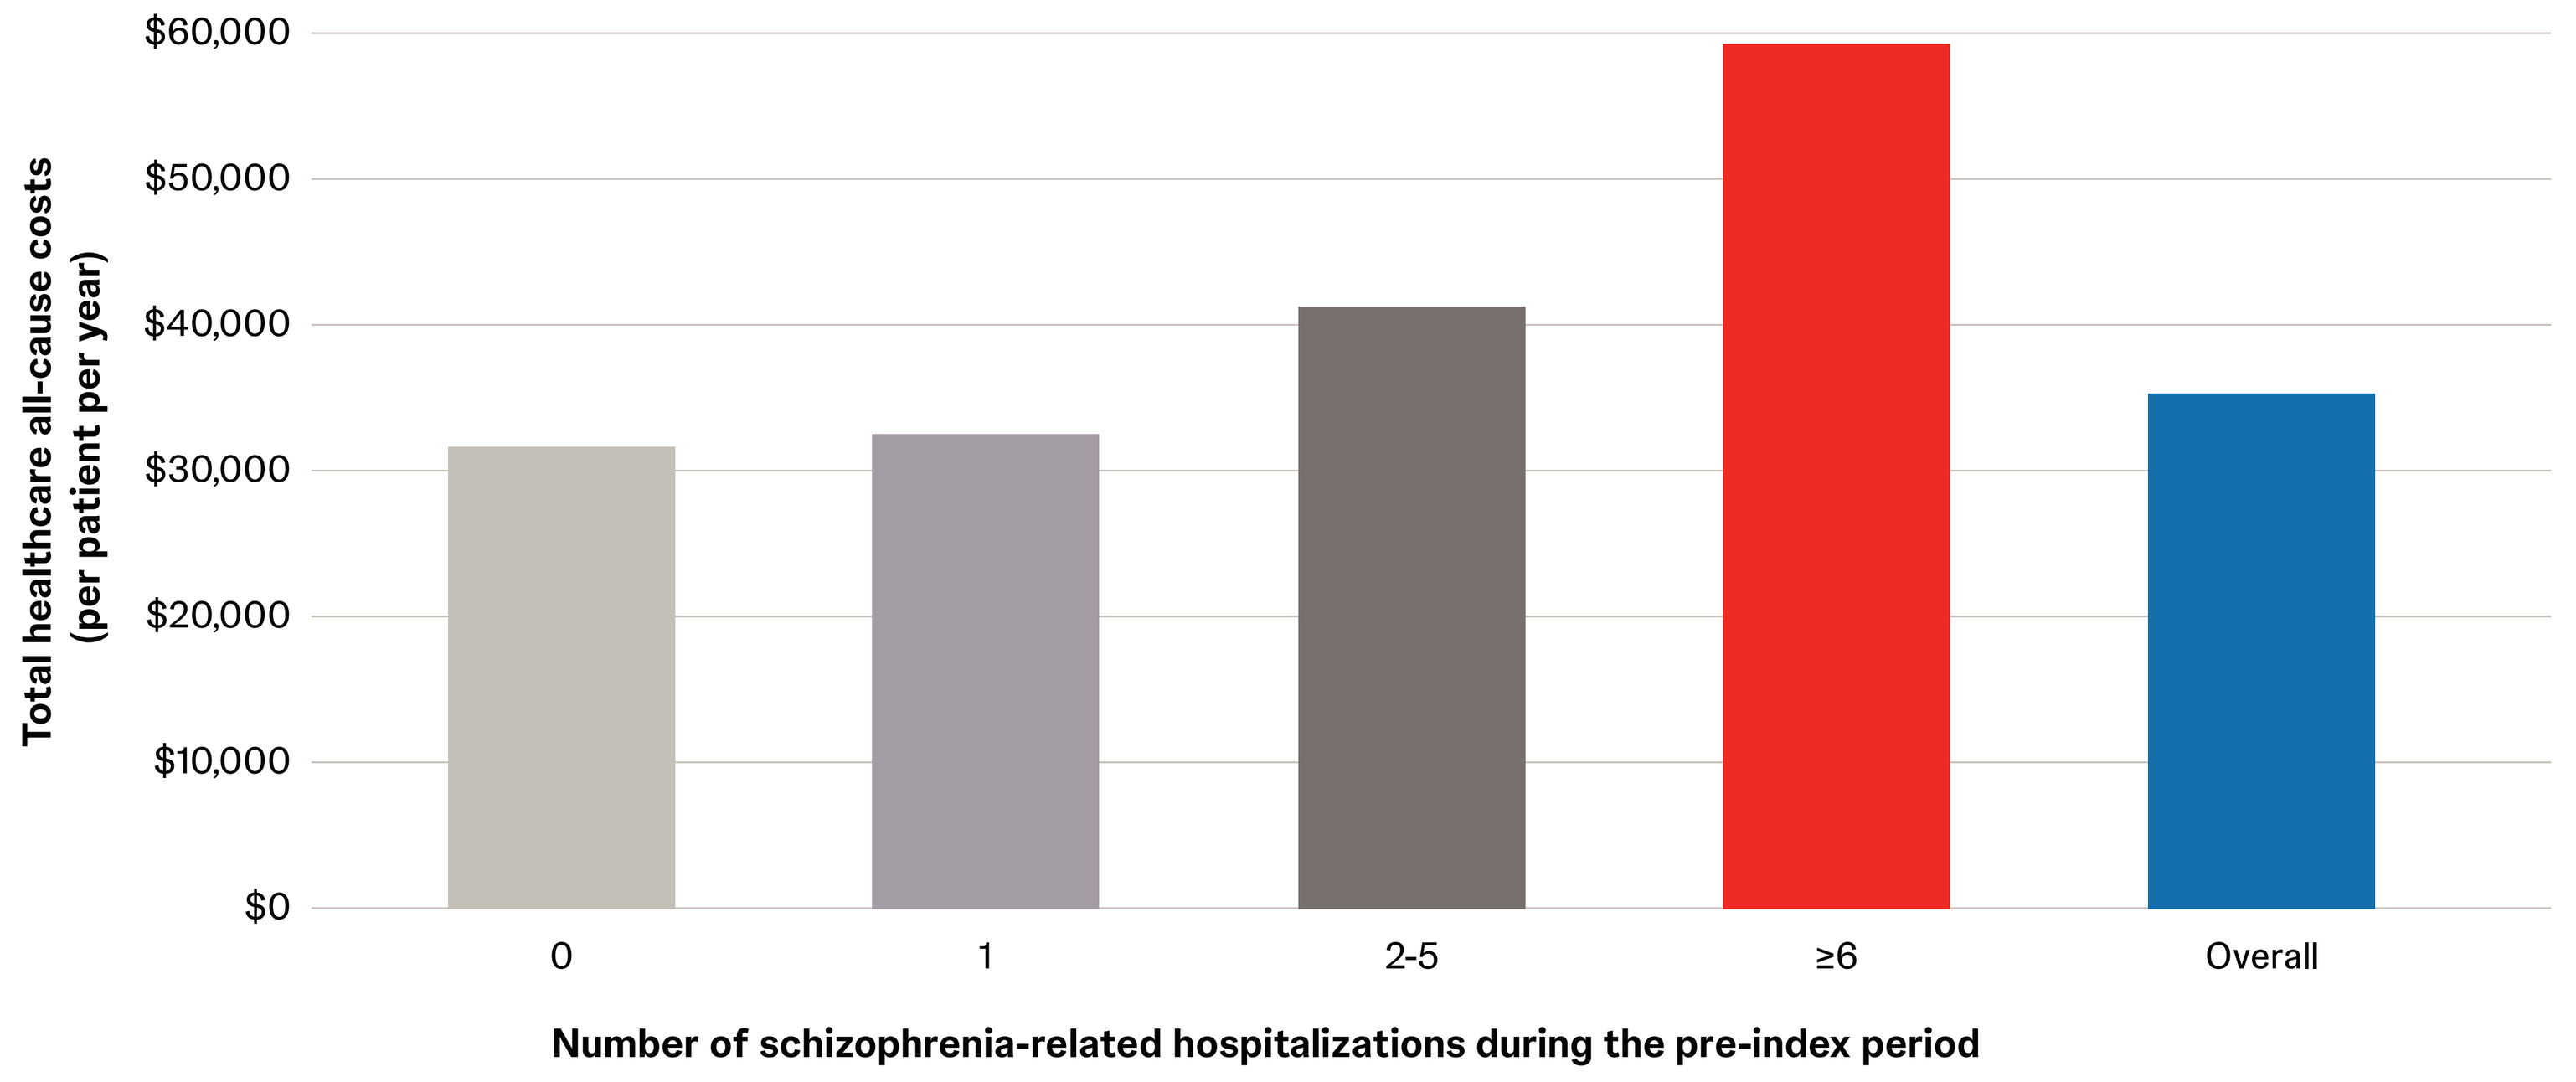
$60,000
$50,000
$40,000
$30,000
$20,000
$10,000
$0
Total healthcare all-cause costs (per patient per year)
0
1
2-5
≥6
Overall
Number of schizophrenia-related hospitalizations during the pre-index period

CX: Add footnote: LAI, long-acting injectable.

Table 4: Linear regression results for continuous outcomes over 12 months following LAI initiation

	Number of schizophrenia-related hospitalizations during the pre-index period			
	0	1	2 to 5	≥6
	Coefficient (95% CI)	Coefficient (95% CI)	Coefficient (95% CI)	Coefficient (95% CI)
Number of hospitalizations				
All-cause hospitalizations	REF	+0.17 (0.01, 0.33)	+0.61 (0.43, 0.79)	+1.89 (1.57, 2.20)
Mental health–related hospitalizations	REF	+0.15 (0.00, 0.30)	+0.58 (0.42, 0.75)	+1.86 (1.57, 2.16)
Schizophrenia-related hospitalization	REF	+0.11 (-0.02, 0.23)	+0.52 (0.38, 0.66)	+1.71 (1.47, 1.96)
Total all-cause costs	REF	+$147.40 (-2812.28, 3107.09)	+$6,324.13 (2,999.25, 9,649.00)	+$20,092.86 (14,197.47, 25,988.26)

LAI, long-acting injectable; REF, reference.

Conclusions

- Young dual-eligible adults with more SCZ-related hospitalizations before LAI initiation had higher hospitalization rates and costs after initiation
- These findings highlight the potential clinical and economic implications of delayed LAI treatment and support efforts to promote earlier initiation to help reduce potentially preventable utilization and downstream spending

Limitations

- Schizophrenia onset cannot be determined precisely in administrative claims due to left censoring and imperfect observability of early symptoms. However, the mean and median ages at which patients had a first observed claim with a schizophrenia diagnosis in our data were consistent with the age of onset reported in the literature
- Adherence and discontinuation measures derived from claims reflect medication availability rather than ingestion, although this limitation is inherent across pharmacoepidemiologic studies
- Relapse was identified using claims-based proxies rather than clinical assessments
- While our analyses adjusted for patient sociodemographic and clinical characteristics available in the claims data, residual confounding may remain, as patients with greater pre-index hospitalizations may differ in unmeasured ways (e.g., symptom severity, social supports, care engagement) that influence both timing of LAI initiation and outcomes

Acknowledgments

Editorial services were provided by ApotheCom, Yardley, PA.

Disclosures

PL reported receiving personal fees from Cobbs Creek Healthcare and SKB Consulting Inc, all unrelated to the submitted work. Ms Benson is an employee of Johnson & Johnson and holds stock in the company. Dr Geng had no disclosures to report. Dr Doshi reported receiving grants from Johnson & Johnson during the conduct of the study; personal fees from AbbVie, Acadia, Janssen, Merck, Otsuka, and Takeda; and grants from Merck and Spark Therapeutics unrelated to the submitted work.

Scan the QR code

Supplementary material are available via QR code.
The QR code is intended to provide scientific information for individual reference, and the information should not be altered or reproduced in any way.

This study was funded by Johnson & Johnson.

Presented at AMCP Nexus 2026; October 26-29, 2026; San Diego, California, USA

References:

1. American Psychiatric Association. *Diagnostic and Statistical Manual of Mental Disorders, Fifth Ed*. American Psychiatric Association Publishing, Inc.; 2013. 2. Marcus SC et al. *J Manag Care Spec Pharm*. 2015;21(9):754-768. 3. El Abdellati K et al. *Front Neurosci*. 2020;14:531763. 4. Olivares JM et al. *Ann Gen Psychiatry*. 2013;12(1):32. 5. Lin D et al. *CNS Drugs*. 2021;35(5):469-481. 6. Li P et al. *Adv Ther*. 2025;42(2):1251-1264. 7. Kane JM et al. *J Clin Psychiatry*. 2023;84(3):22m14544.

This material is distributed for scientific purposes by Johnson & Johnson Innovative Medicine and is not for promotional use.