44

Longitudinal Real-World Study of the Patient Journey of a National Sample of Beneficiaries With Schizophrenia Dually Eligible for Medicare & Medicaid

Pengxiang Li[1], Zhi Geng[1], Carmela Benson[2], Charmi Patel[2], Jalpa A. Doshi[1]

[1]University of Pennsylvania, Philadelphia, PA; [2]Janssen Scientific Affairs, LLC, a Johnson & Johnson company, Titusville, NJ

Introduction

- Schizophrenia (SCZ) is a chronic mental illness characterized by delusions, hallucinations, and other disabling psychiatric symptoms which affects approximately 1% of the population in the US[1,2]
- Antipsychotics (APs) are the mainstay therapy for the management of SCZ
 - Adherence is critical to improve disease symptoms, reduce risk of acute psychotic episodes, and decrease the frequency of relapses and psychiatric hospitalizations
 - Historically, daily oral antipsychotic use has been the mainstay of schizophrenia treatment; however, its effectiveness is hindered by poor adherence
- Long-acting injectable antipsychotics (LAIs) provide an important treatment option for patients who struggle with adherence since they need to be administered less frequently than the daily use of oral agents
 - Despite their benefits, utilization of LAIs in clinical practice has been relatively low and the majority of patients, including those who have a history of relapse, continue to receive oral antipsychotics (OAP)
- To understand the mismatch between LAI performance vs utilization, assessment of the patient journey from the time of SCZ diagnosis to initiation of first antipsychotic treatment is imperative
 - Prior studies have been based on individual segments of the journey and come from disparate and older (10+ years) data sources
 - No prior study has examined this topic in the U.S. Medicare and Medicaid population, despite the fact that about half of all patients receive coverage via Medicare and the majority of these patients are dually eligible for Medicaid

Objective

- To describe the real-world patient journey before LAI initiation among Medicare-Medicaid enrollees (i.e., dual eligibles) with SCZ using longitudinal national data

Methods

Data source

- This study linked two data sources available from the Centers for Medicare and Medicaid Services
- 2006 to 2021 Medicare Chronic Conditions Data Warehouse (CCW) 100% fee-for-service (FFS) data files
- 2006 to 2021 national Medicaid fee-for-service or managed care data files
 - 2006 to 2015 Medicaid Analytic eXtract (MAX) files
 - 2014 to 2021 Transformed Medicaid Statistical Information System Analytic 2iles (TAF) files

Study design and sample

- This was a retrospective longitudinal cohort study
- The sample included all fee-for-service disabled dually-eligible Medicare beneficiaries with SCZ and ≥1 antipsychotic fill between 01/01/2009 to 12/31/2018. Patients were required to meet the following selection criteria:
 - First date of SCZ diagnosis in Medicare or Medicaid claim files between 2009 and 2018
 - First eligible for Medicare between 2009 and 2018
 - Reason for Medicare eligibility was disability rather than age ≥65 (given these patients are more likely to be earlier in their patient journey since initial SCZ diagnosis)
 - Evidence of dually eligibility in the first year of Medicare eligibility
 - For patients with first SCZ diagnosis under Medicaid:
 - Continuous full Medicaid coverage between first SCZ diagnosis date and initial Medicare eligibility date, and
 - Continuous fee-for-service Medicare Parts A, B, and D coverage from first Medicare eligibility date until death, entry into Medicare Advantage, or end of follow-up (i.e. 12/31/2021)
 - For patients with first SCZ diagnosis under Medicare:
 - Continuous fee-for-service Medicare Parts A, B, and D coverage from first SCZ diagnosis date until death, entry into Medicare Advantage, or end of follow-up (i.e. 12/31/2021)
 - First AP fill date occurred on or after the date of the first observed SCZ diagnosis date in Medicare or Medicaid claims and before the end of follow-up
- The overall sample was further classified into seven non-mutually exclusive samples of new initiators of any LAI, any first-generation LAI (FGA LAIs), any second-generation LAI (SGA LAIs), risperidone LAI, paliperidone once-monthly (PP1M) LAI, paliperidone every 3 months (PP3M) LAI, and aripiprazole LAI

Outcomes

- Age at first schizophrenia diagnosis and LAI initiation date
- Treatment utilization prior to LAI initiation
 - Time to OAP initiation
 - Mean (SD) number of OAP agents
 - Type of OAP agents tried (top 10 most commonly used agents)
- Evidence of relapse
 - Relapse was defined as claims with a schizophrenia diagnosis in first or second diagnosis position of inpatient or emergency room (ER) claims
- Schizophrenia-related ER visits and hospitalizations were defined as those with a diagnosis code for schizophrenia in the primary or secondary positions on the index date
 - Mean (SD) number of visits/hospitalizations
 - Mean (SD) costs per visit/hospitalization
- LAI treatment initiation
 - Time to first LAI treatment
 - Type of LAI treatment initiated first and at any point over follow-up

Analysis

- Descriptive analyses were conducted for all study outcomes
- All analyses were conducted using SAS Enterprise Guide 9.4 version

Results

- Over a median follow-up of 5.6 years from AP initiation, only 36% of the 25,356 patients in our sample received LAIs (**Figure 1**)
- Before initiating LAIs, patients cycled through multiple OAPs (45% tried 1 or 2; 27% tried 3 or more OAP agents, **Figure 1**)
 - PP1M LAI initiators had tried several different oral agents whereas most other LAI initiators had largely tried the same oral molecule (e.g. aripiprazole oral to aripiprazole LAI, **Table 1**)
- 63% of patients had evidence of relapse (SCZ-related ER or inpatient visit) prior to LAI initiation (**Figure 1**)
 - The relapse rate prior to LAI initiation ranged from 58.9% (risperidone LAI) to 79.8% (PP3M LAI) (**Table 1**)
- 43% of patients had evidence of SCZ-related ER visits prior to LAI initiation (**Figure 1**)
 - Mean (SD) number of visits: 3.5 (18.7)
 - The mean (SD) number of visits ranged from 2.0 (8.2) in the risperidone LAI group to 10.7 (39.9) in the PP3M LAI group (**Table 1**)
 - Mean (SD) cost per visit: $328 ($285)
 - Costs per visit were generally similar across groups
- 50% of patients had evidence of SCZ-related hospitalization prior to LAI initiation (**Figure 1**)
 - Mean (SD) number of hospitalizations: 1.4 (2.5)
 - The mean (SD) number of hospitalizations ranged from 1.5 (3.2) in the risperidone LAI group to 2.6 (4.5) in the aripiprazole LAI group (**Table 1**)
 - Mean (SD) cost per hospitalizations: $10,488 ($11,024)
 - Costs per visit were generally similar across groups

Figure: Patient journey prior to LAI initiation among disabled dually eligible Medicare beneficiaries who initiated an LAI

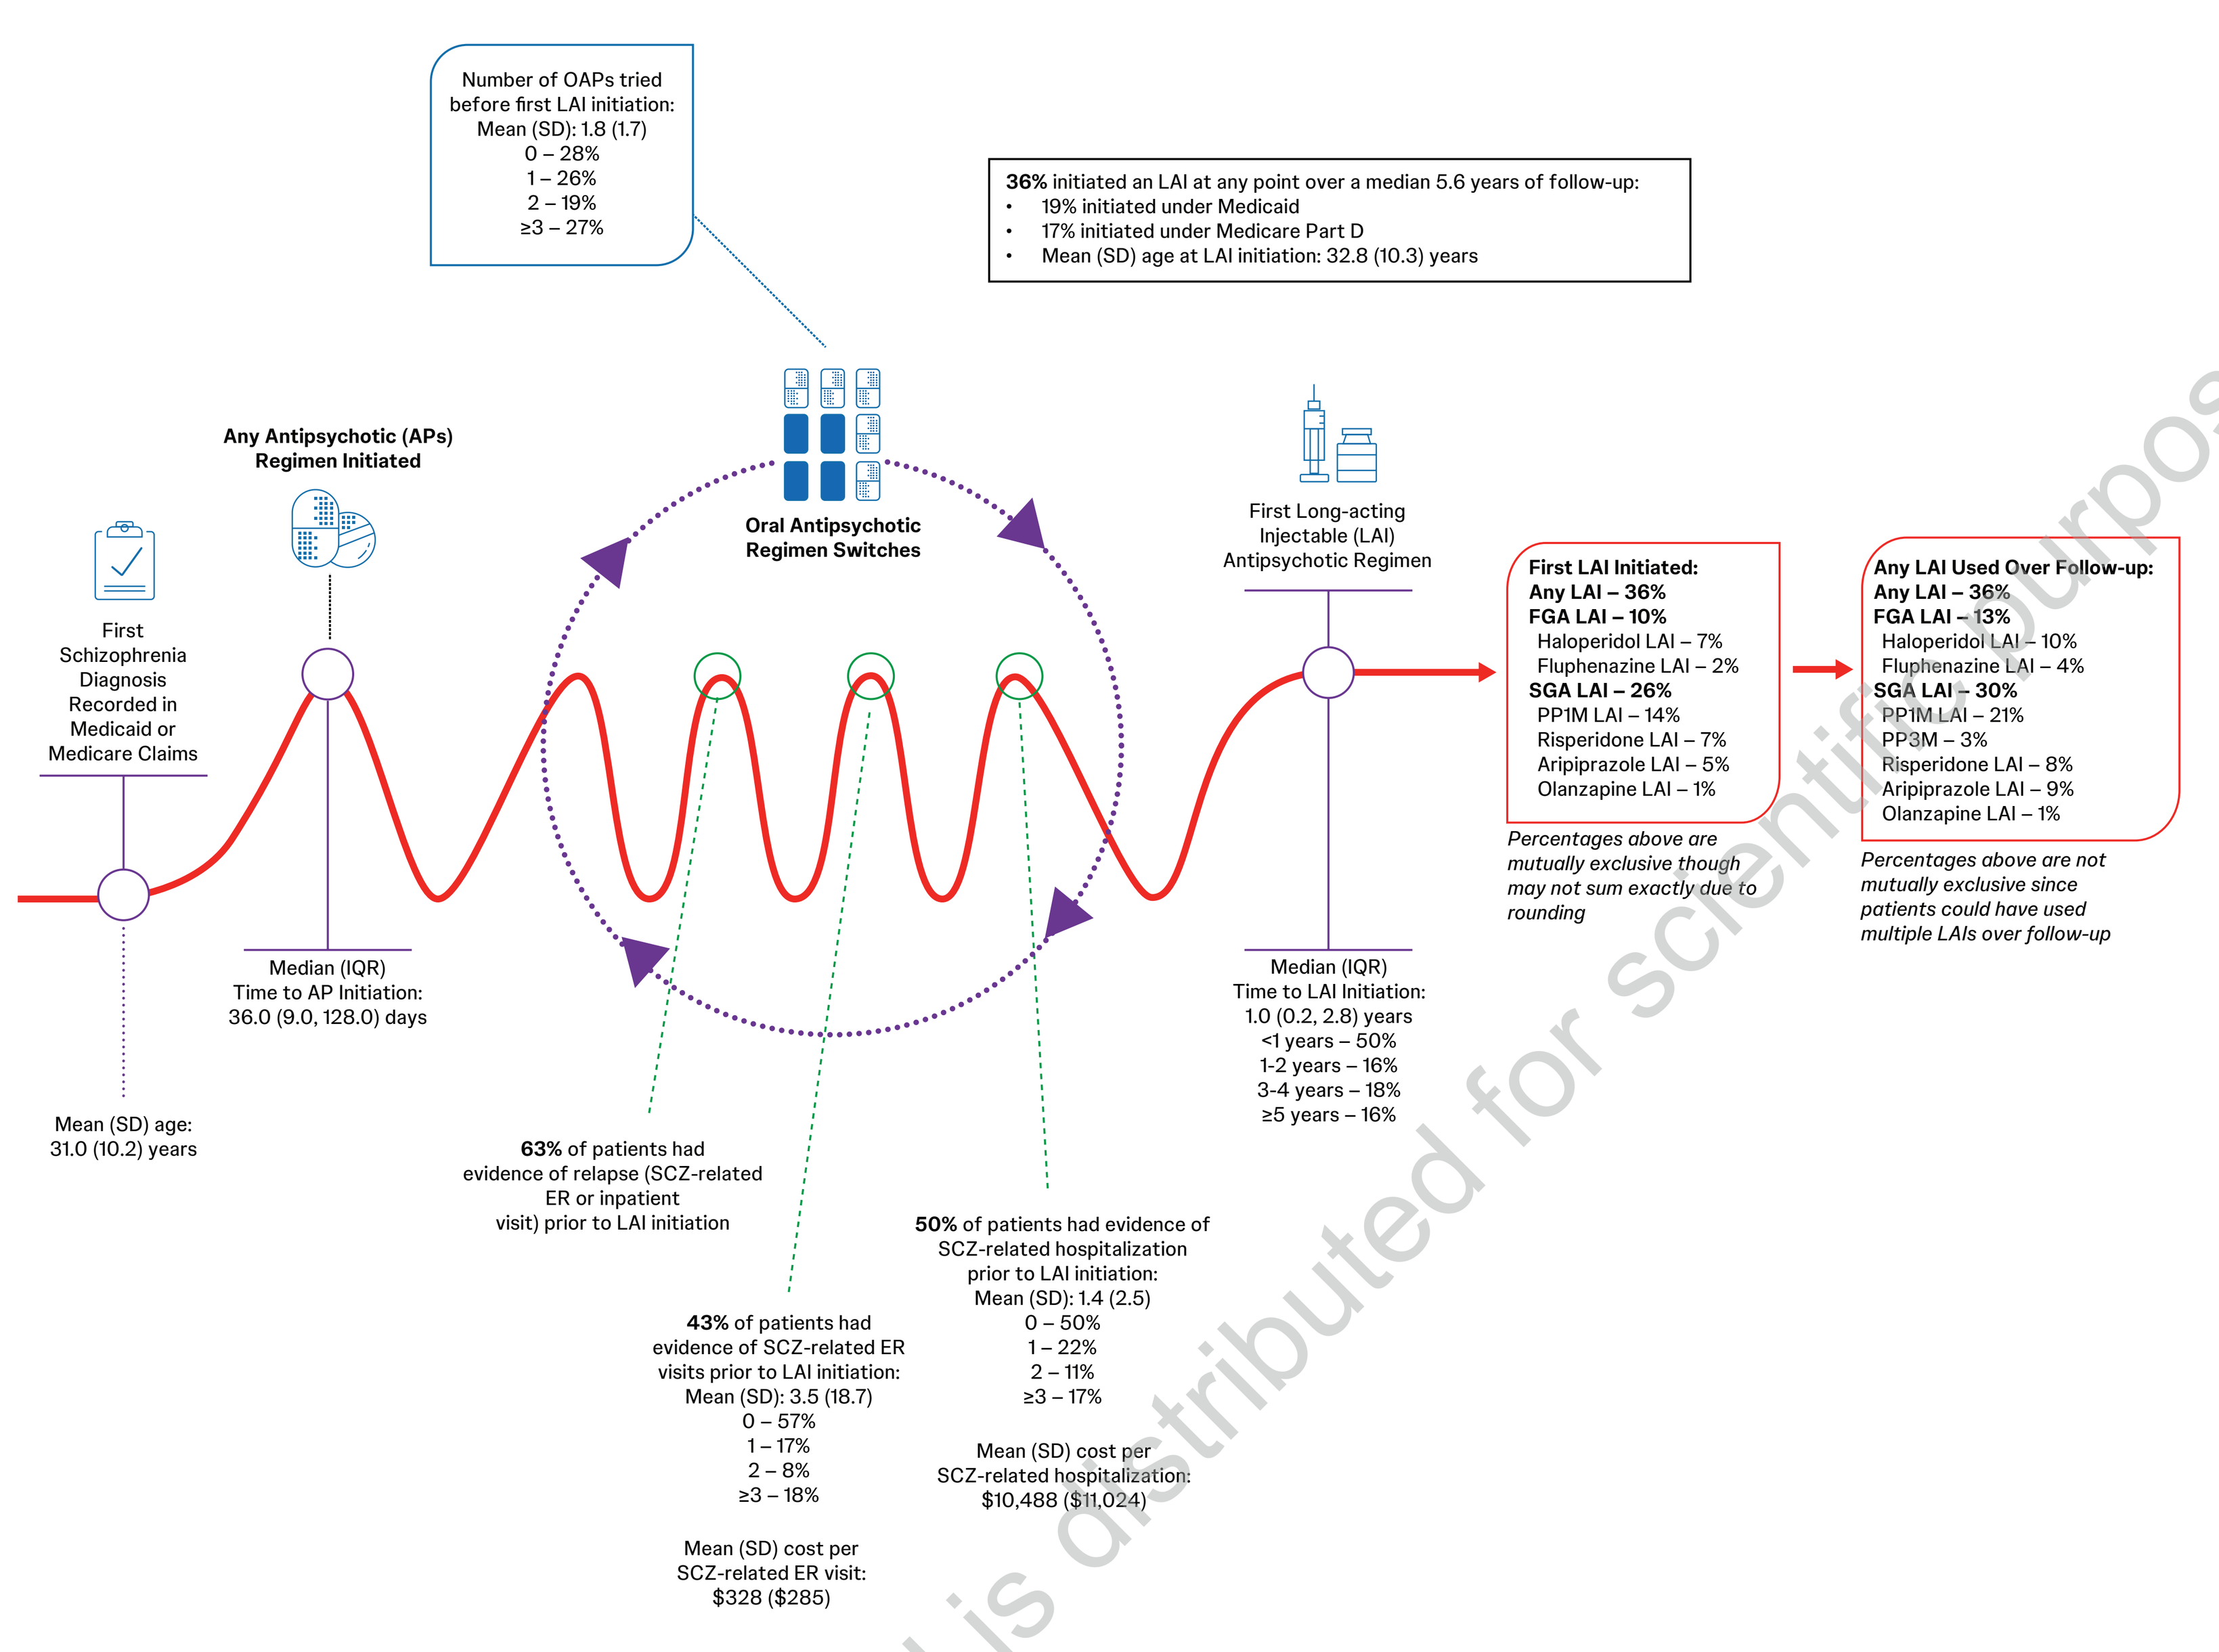
Number of OAPs tried before first LAI initiation: Mean (SD): 1.8 (1.7)
0 – 28%
1 – 26%
2 – 19%
≥3 – 27%
36% initiated an LAI at any point over a median 5.6 years of follow-up:
19% initiated under Medicaid
17% initiated under Medicare Part D
Mean (SD) age at LAI initiation: 32.8 (10.3) years
First Schizophrenia Diagnosis Recorded in Medicaid or Medicare Claims
Any Antipsychotic (APs) Regimen Initiated
Oral Antipsychotic Regimen Switches
First Long-acting Injectable (LAI) Antipsychotic Regimen
First LAI Initiated:
Any LAI – 36%
FGA LAI – 10%
Haloperidol LAI – 7%
Fluphenazine LAI – 2%
SGA LAI – 26%
PP1M LAI – 14%
Risperidone LAI – 7%
Aripiprazole LAI – 5%
Olanzapine LAI – 1%
Percentages above are mutually exclusive though may not sum exactly due to rounding
Any LAI Used Over Follow-up:
Any LAI – 36%
FGA LAI – 13%
Haloperidol LAI – 10%
Fluphenazine LAI – 4%
SGA LAI – 30%
PP1M LAI – 21%
PP3M – 3%
Risperidone LAI – 8%
Aripiprazole LAI – 9%
Olanzapine LAI – 1%
Percentages above are not mutually exclusive since patients could have used multiple LAIs over follow-up
Mean (SD) age: 31.0 (10.2) years
Median (IQR) Time to AP Initiation: 36.0 (9.0, 128.0) days
63% of patients had evidence of relapse (SCZ-related ER or inpatient visit) prior to LAI initiation
43% of patients had evidence of SCZ-related ER visits prior to LAI initiation: Mean (SD): 3.5 (18.7)
0 – 57%
1 – 17%
2 – 8%
≥3 – 18%
Mean (SD) cost per SCZ-related ER visit: $328 ($285)
50% of patients had evidence of SCZ-related hospitalization prior to LAI initiation: Mean (SD): 1.4 (2.5)
0 – 50%
1 – 22%
2 – 11%
≥3 – 17%
Mean (SD) cost per SCZ-related hospitalization: $10,488 ($11,024)
Median (IQR) Time to LAI Initiation: 1.0 (0.2, 2.8) years
<1 years – 50%
1-2 years – 16%
3-4 years – 18%
≥5 years – 16%

AP, antipsychotic; ER, emergency room; FGA, first generation antipsychotic; IQR, interquartile range; LAI, long acting injectable; OAP, oral antipsychotic; PP1M, paliperidone palmitate once monthly; PP3M, paliperidone palmitate once every 3 months; SGA, second generation antipsychotic; SCZ, schizophrenia.

Table: Antipsychotic treatment utilization, relapse, and schizophrenia-related ER visits and hospitalizations prior to initiation of the specific LAI

	Index LAI						
			SGA LAI				
	Any LAI (N=9,107)	Any FGA LAI (N=3,223)	Any SGA LAI (N=7,558)	Risperidone LAI (N=2,110)	PP1M LAI (N=5,226)	PP3M LAI (N=881)	Aripiprazole LAI (N=2,173)
Age on first SCZ diagnosis date recorded in Medicaid or Medicare claims, mean (SD)	31.0 (10.2)	30.9 (10.2)	30.6 (9.9)	30.8 (10.1)	30.2 (9.6)	30.3 (9.5)	30.0 (9.4)
≤24 years	31.4%	32.3%	32.7%	32.3%	33.9%	30.5%	32.7%
25 to 34 years	40.6%	40.0%	40.9%	40.6%	41.5%	44.9%	42.6%
35 to 44 years	14.9%	14.9%	14.5%	15.5%	13.7%	12.7%	13.8%
45 to 54 years	9.3%	8.9%	8.7%	7.8%	8.2%	8.1%	7.7%
≥55 years	3.8%	4.0%	3.2%	3.7%	2.8%	2.8%	2.5%
Days from first SCZ diagnosis to any AP initiation, median (IQR)	36.0 (9.0, 128.0)	32.0 (9.0, 117.0)	37.0 (9.0, 132.0)	30.0 (8.0, 109.0)	39.0 (10.0, 131.0)	41.0 (8.0, 132.0)	37.0 (9.0, 141.0)
Years from first SCZ diagnosis in Medicaid or Medicare claims to index LAI initiation date, median (IQR)	1.0 (0.2, 2.8)	1.3 (0.2, 3.3)	1.3 (0.3, 3.4)	0.8 (0.1, 2.4)	1.8 (0.6, 3.9)	4.7 (2.6, 6.6)	3.3 (1.4, 5.4)
<1 years	49.9%	44.8%	43.3%	53.9%	35.3%	7.0%	19.1%
1-2 years	16.2%	16.5%	16.5%	16.4%	17.0%	11.1%	14.1%
3-4 years	18.2%	18.7%	20.4%	18.1%	23.9%	23.5%	25.0%
≥5 years	15.7%	19.9%	19.9%	11.6%	23.7%	58.3%	41.7%
Age on index LAI initiation date, mean (SD)	32.8 (10.3)	33.0 (10.1)	32.7 (10.2)	32.3 (10.3)	32.6 (9.9)	34.9 (9.8)	33.6 (9.7)
≤24 years*	21.6%	19.7%	21.1%	23.4%	20.1%	6.9%	14.7%
25 to 34 years	45.2%	46.3%	46.2%	45.3%	47.6%	54.1%	49.9%
35 to 44 years	17.9%	19.4%	18.0%	17.7%	18.2%	22.7%	20.9%
45 to 54 years	10.2%	9.4%	10.1%	9.1%	10.0%	10.0%	9.5%
≥55 years	5.1%	5.3%	4.7%	4.6%	4.2%	6.2%	5.0%
Treatment utilization between first SCZ diagnosis date and index LAI initiation							
Mean (SD) number of different OAP agents tried	1.8 (1.7)	2.1 (2.0)	2.0 (1.9)	1.7 (1.8)	2.2 (1.9)	2.7 (2.1)	3.0 (2.1)
Tried 0 OAPs before LAI	27.9%	25.2%	24.1%	30.4%	20.5%	12.5%	9.6%
Tried 1 OAP before LAI	25.7%	22.7%	24.5%	26.3%	23.3%	20.7%	17.2%
Tried 2 OAPs before LAI	19.4%	18.5%	20.0%	18.1%	20.6%	18.7%	21.4%
Tried 3 or more OAPs before LAI	27.0%	33.6%	31.5%	25.3%	35.6%	48.1%	51.9%
Type of OAP agents tried (top 10 most commonly used OAPs)							
Any risperidone OAP	34.3%	31.6%	39.2%	54.8%	44.3%	50.7%	40.4%
Any olanzapine OAP	23.8%	26.2%	26.5%	20.0%	29.7%	34.7%	35.2%
Any quetiapine OAP	21.6%	23.6%	23.5%	18.1%	25.3%	29.2%	32.6%
Any aripiprazole OAP	21.3%	18.9%	25.3%	14.9%	21.2%	25.1%	66.3%
Any haloperidol OAP	19.8%	40.6%	19.2%	16.0%	21.5%	26.8%	27.8%
Any paliperidone OAP	10.6%	9.2%	13.5%	6.7%	20.1%	36.8%	15.6%
Any fluphenazine OAP	5.3%	13.1%	5.6%	5.5%	6.4%	9.2%	8.7%
Any clozapine OAP	5.1%	7.7%	5.4%	4.7%	6.2%	8.3%	8.4%
Any ziprasidone OAP	9.4%	10.0%	10.3%	8.4%	11.1%	11.7%	14.3%
Any lurasidone OAP	8.8%	8.9%	10.0%	4.9%	11.2%	14.0%	17.9%
Evidence of relapse between first SCZ diagnosis date and index LAI initiation	63.4%	69.2%	66.3%	58.9%	70.7%	79.8%	78.0%
SCZ-related ER visit between first SCZ diagnosis date and index LAI initiation	42.9%	48.6%	46.5%	40.5%	51.2%	63.6%	59.6%
Number of SCZ-related ER visits, mean (SD)	3.5 (18.7)	4.8 (21.1)	4.1 (20.4)	2.0 (8.2)	5.4 (24.5)	10.7 (39.9)	7.3 (25.1)
0 SCZ-related ER visits	57.1%	51.4%	53.5%	59.5%	48.8%	36.4%	40.4%
1 SCZ-related ER visit	16.6%	15.9%	16.9%	17.4%	17.2%	17.3%	16.9%
2 SCZ-related ER visits	8.0%	8.5%	8.7%	8.2%	9.2%	9.8%	9.4%
3 or more SCZ-related ER visits	18.2%	24.2%	20.8%	14.8%	24.8%	36.5%	33.2%
Cost per SCZ-related ER visit, mean (SD)**	$328 ($285)	$318 ($244)	$331 ($282)	$290 ($225)	$321 ($249)	$335 ($293)	$373 ($348)
SCZ-related hospitalization between first SCZ diagnosis date and index LAI initiation	49.7%	59.0%	52.1%	47.6%	57.2%	61.9%	62.0%
Number of SCZ-related hospitalizations, mean (SD)	1.4 (2.5)	2.2 (3.9)	1.6 (3.0)	1.5 (3.2)	2.0 (3.6)	2.3 (3.8)	2.6 (4.5)
0 SCZ-related hospitalizations	50.3%	41.0%	47.9%	52.4%	42.8%	38.1%	38.0%
1 SCZ-related hospitalization	21.8%	20.3%	21.0%	19.9%	20.4%	19.9%	20.1%
2 SCZ-related hospitalizations	10.9%	10.9%	11.5%	10.5%	12.4%	14.0%	11.6%
3 or more SCZ-related hospitalizations	17.0%	27.8%	19.6%	17.3%	24.4%	28.0%	30.4%
Cost per SCZ-related hospitalization, mean (SD)**	$10,488 ($11,024)	$11,175 ($12,128)	$10,410 ($10,143)	$9,929 ($8,550)	$10,239 ($9,479)	$10,584 ($8,762)	$10,676 ($10,363)

AP, antipsychotic; ER, emergency room; IQR, interquartile range; LAI, long acting injectable; OAP, oral antipsychotic; PP1M, paliperidone palmitate once monthly; PP3M, paliperidone palmitate once every 3 months; SCZ, schizophrenia.
*The number of patients aged younger than 18 years was less than 11 in any of the LAI groups and cannot be reported per CMS' Cell Size Suppression Policy.
**Mean cost calculated only from fee-for-service Medicaid and Medicare claims since cost information in Medicaid managed care encounter records is unreliable.

Key Takeaways

- Despite their availability and treatment guideline recommendations, only about 1 in 3 Medicare beneficiaries initiated an LAI over a period of more than 5 years
- It is still common for patients to cycle through multiple oral agents before initiating an LAI, but there is a shift to early initiation of LAIs as soon as patients become eligible for Medicare coverage
- High rates of relapse, emergency room visits, and hospitalizations may lead to LAI initiation

Conclusions

- This is the first study to describe the real-world journey of patients with SCZ before LAI initiation using national long-term follow-up data on dually eligible Medicare and Medicaid beneficiaries
- Only one-third of dual-eligible patients with SCZ received an LAI over a median follow-up of 5.6 years after their first recorded SCZ diagnosis in Medicare or Medicaid claims
- Prior to LAI initiation, patients cycled through multiple oral antipsychotic treatments; more than 1 in 4 patients tried 3 or more oral antipsychotics before initiating an LAI
- Patients experienced high rates of relapse, hospitalizations, and ER visits before they were initiated on an LAI
- Our findings should serve as a call to action for mental health practitioners and policymakers to increase efforts to improve quality of care and outcomes in this vulnerable population

Limitations

- This was a descriptive study, and hence no causal conclusions can be drawn
- Findings are only generalizable to beneficiaries dually eligible for Medicare and Medicaid and not to those with other sources of or lack of insurance coverage
- LAIs received in an inpatient setting may not have been recorded as a prescription, and as such were not captured in this analysis

Disclosures
PL reported receiving personal fees from Avalon Health Economics, LLC; HealthStatistics, LLC; SKB Consulting Inc; and Cobbs Creek Healthcare, all unrelated to the submitted work. CB and CP reported being employees of Janssen Scientific Affairs, LLC, and stockholders of Johnson and Johnson. JAD reported receiving grants from Janssen Scientific Affairs, LLC, during the conduct of the study; personal fees from AbbVie, Acadia, Janssen, Merck, Otsuka, and Takeda; and grants from Merck and Spark Therapeutics unrelated to the submitted work. No other authors had disclosures to report.

Neuropsychiatry

Scan the QR code

The QR code is intended to provide scientific information for individual reference, and the information should not be altered or reproduced in any way.

This study was funded by Janssen Scientific Affairs, LLC, a Johnson & Johnson company.

Presented at 2025 Psych Congress Elevate; May 28-31, 2025; Las Vegas, Nevada, USA

References

1. American Psychiatric Association. Diagnostic and Statistical Manual of Mental Disorders: DSM-V. 5th ed. Washington, DC: American Psychiatric Association Publishing; 2013. 2. Regier DA, Narrow WE, Rae DS, et al. The de facto US mental and addictive disorders service system: Epidemiologic Catchment Area prospective 1-year prevalence rates of disorders and services. Archives of General Psychiatry. 1993;50(2):85-94.

This material is distributed for scientific purposes on J&J Medical Connect, and is not for promotional use