Long-Term Efficacy and Safety of Esketamine Nasal Spray as Monotherapy: A Post Hoc Analysis of the SUSTAIN-3 Study

John Zajecka[1], Dong-Jing Fu[2], Ibrahim Turkoz[2], Patricia Cabrera[2], Oliver Lopena[2], Lisa Lim[2], Megan Kunka Fritz[2], Muhammad Ahmed[2]

[1]Rush University Medical Center, Chicago, IL; [2]Johnson & Johnson, Titusville, NJ

Introduction

- Prior to January 2025, esketamine nasal spray (ESK) was approved for treatment-resistant depression (TRD) in adults and for depressive symptoms in adults with major depressive disorder with acute suicidal ideation or behavior, both in conjunction with an oral antidepressant (OAD)[1]
- Primary findings from a phase 4, multicenter, double-blind, randomized, placebo-controlled trial (TRD4005, NCT04599855) demonstrated that ESK as monotherapy led to a rapid and superior improvement in Montgomery-Åsberg Depression Rating Scale (MADRS) total score compared with placebo at day 28, and as early as 24 hours after the first dose[2]
 - Based on these results, ESK was approved in the United States as the first and only monotherapy for adults with TRD[1,3]
- Some patients enrolled in SUSTAIN-3, a long-term open-label ESK efficacy and safety study, received ESK treatment without a concurrent OAD for a prolonged period (≥12 consecutive months) at the investigators' discretion[4,5]
 - Data from these patients can provide additional evidence to inform the long-term efficacy and safety of ESK monotherapy

Objective

- To describe the long-term efficacy and safety of ESK in adults with TRD who received ESK as monotherapy for ≥12 consecutive months in a long-term open-label study

Methods

Study design

- SUSTAIN-3 was a phase 3, open-label, long-term extension study comprised of 2 phases: a 4-week induction phase (IND) and a variable duration optimization/maintenance phase (OP/M) (**Figure 1**)
 - SUSTAIN-3 enrolled patients with TRD (≥2 prior OAD treatment failures) from 1 of 6 parent studies
 - Patients entered either the IND or OP/M phase, based on their status at the end of the parent study
- Patients in SUSTAIN-3 received open-label, flexibly-dosed ESK, with a concurrent OAD; however, this post hoc subgroup analysis included patients who either never received an OAD or stopped OAD treatment but continued with ESK for variable durations
 - Patients who discontinued OAD treatment may have reinitiated OAD treatment at a later point, but received ESK monotherapy for ≥12 consecutive months during the study period
- During the IND phase, ESK was flexibly-dosed twice weekly for 4 weeks; for patients who proceeded to the OP/M phase, ESK dosing frequency was individualized based on depression severity and tolerability to weekly, every other week, or every 4 weeks[1]

Assessments

- Efficacy outcomes included changes in disease severity, evaluated per the MADRS and the 9-item Patient Health Questionnaire (PHQ-9)
- The proportions of patients who attained remission status were determined
 - For MADRS, remission was defined as a total score ≤12
 - For PHQ-9, remission was defined as a total score <5
- Treatment-emergent adverse events (TEAEs) were monitored throughout the duration of the study

Figure 1: Study design

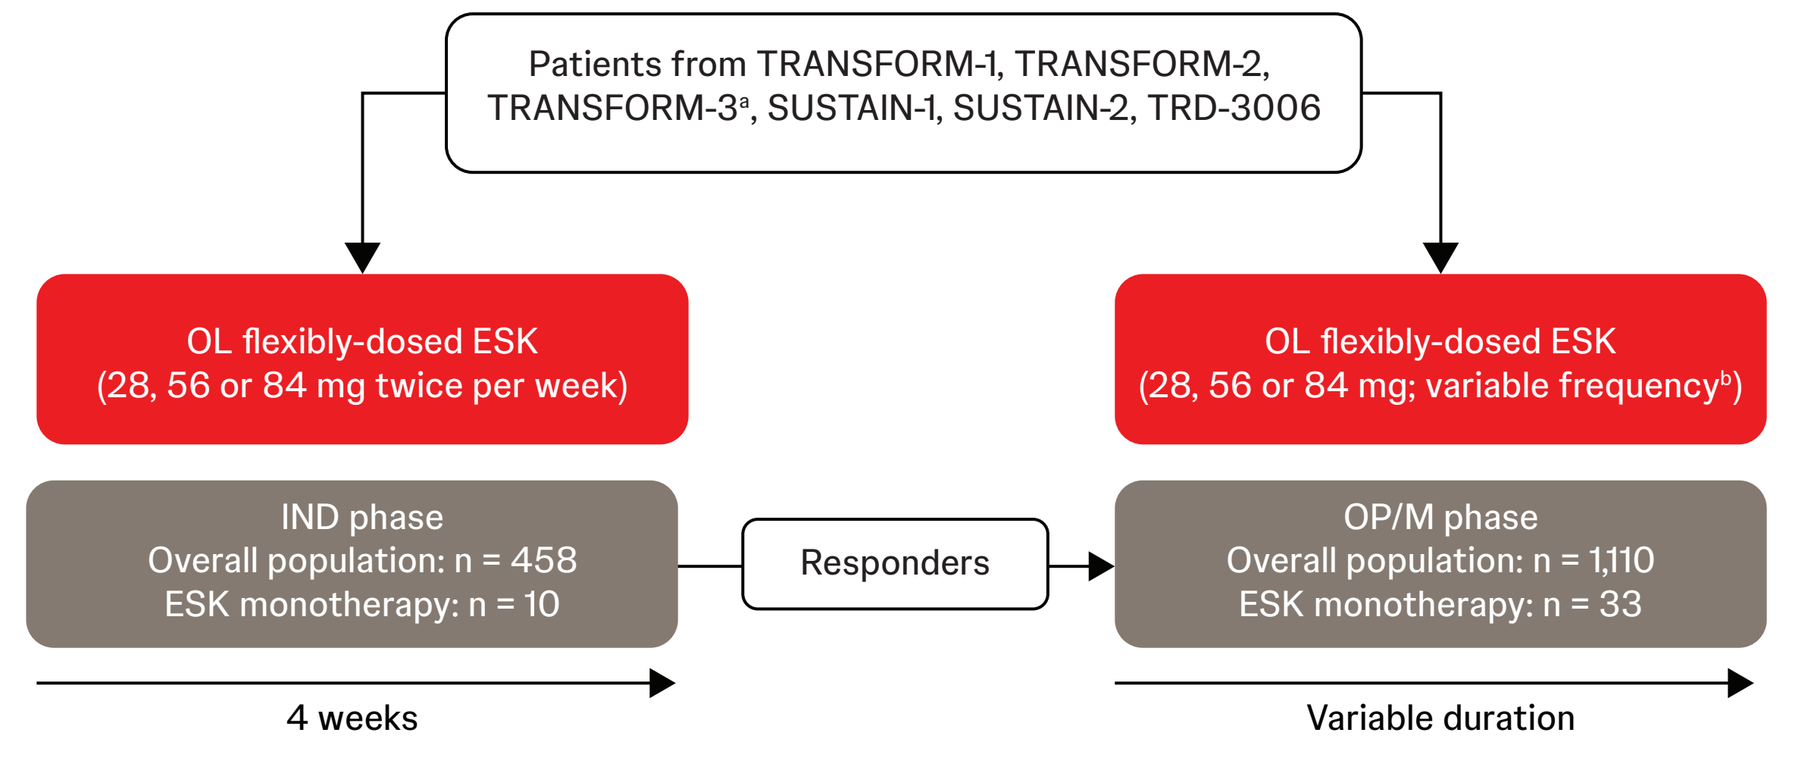
Patients from TRANSFORM-1, TRANSFORM-2, TRANSFORM-3ᵃ, SUSTAIN-1, SUSTAIN-2, TRD-3006
OL flexibly-dosed ESK (28, 56 or 84 mg twice per week)
OL flexibly-dosed ESK (28, 56 or 84 mg; variable frequencyᵇ)
IND phase
Overall population: n = 458
ESK monotherapy: n = 10
Responders
OP/M phase
Overall population: n = 1,110
ESK monotherapy: n = 33
4 weeks
Variable duration

ESK, esketamine nasal spray; IND, induction; OAD, oral antidepressant; OL, open-label; OP/M, optimization/maintenance.
ESK monotherapy patients were defined as patients who did not receive OADs for ≥12 consecutive months at any point during ESK treatment.
[a]Results from the TRANSFORM-3 study (patients aged ≥65 years) were not included in this subgroup analysis.
[b]Dosing frequency adjusted based on Clinical Global Impression-Severity scale score and tolerability. Dosing frequency during OP/M could be weekly, every other week, or every 4 weeks.

Table 1: Baseline characteristics

	ESK monotherapy patients N = 33	Overall SUSTAIN-3 patients N = 1,148
Mean age (SD), years	44.5 (11.27)	49.6 (12.28)
Sex, n (%)		
Female	18 (54.5)	764 (66.6)
Male	15 (45.5)	384 (33.4)
Race, n (%)		
American Indian or Alaskan Native	0	1 (0.1)
Asian	4 (12.1)	45 (3.9)
Black or African American	2 (6.1)	45 (3.9)
White	25 (75.8)	996 (86.8)
Other	1 (3.0)	29 (2.5)
Multiple	0	10 (0.9)
Not reported	1 (3.0)	22 (1.9)
Mean duration of current MDD episode (SD), weeks	197.9 (229.95)	162.8 (257.21)
Mean (SD) baseline MADRS total score[a]		
IND baseline	33.8 (3.82)[b]	29.1 (7.96)[d]
OP/M baseline	11.2 (9.51)[c]	13.1 (8.79)[e]
Mean (SD) baseline PHQ-9 total score[a]		
IND baseline	17.2 (4.39)[b]	15.4 (5.59)[d]
OP/M baseline	6.0 (4.82)[c]	7.6 (5.60)[e]

ESK, esketamine nasal spray; IND, induction phase; MADRS, Montgomery-Åsberg Depression Rating Scale; MDD, major depressive disorder; OAD, oral antidepressant; OP/M, optimization/maintenance phase; PHQ-9, 9-item Patient Health Questionnaire.
ESK monotherapy patients were defined as patients who did not receive OADs for ≥12 consecutive months at any point during ESK treatment.
[a]Patients could enter SUSTAIN-3 at either the IND phase or OP/M phase, based on their status at the end of the parent study. [b]N = 10. [c]N = 33. [d]N = 458. [e]N = 1,110.

Table 2: Most common TEAEs

TEAE, n (%)	ESK monotherapy patients N = 33
Dizziness	18 (54.6)
Headache	17 (51.5)
Nausea	13 (39.4)
Upper respiratory tract infection	12 (36.4)
Back pain	9 (27.3)
Diarrhea	9 (27.3)
Anxiety	8 (24.2)
Dissociation	8 (24.2)
Arthralgia	7 (21.2)
Dysgeusia	7 (21.2)
Fatigue	7 (21.2)
Insomnia	7 (21.2)

ESK, esketamine nasal spray; OAD, oral antidepressant; TEAE, treatment-emergent adverse event.
ESK monotherapy patients were defined as patients who did not receive OADs for ≥12 consecutive months at any point during ESK treatment.

Results

Baseline characteristics

- A total of 1,148 patients were enrolled in SUSTAIN-3; this subgroup analysis included 33 patients who received ESK monotherapy for ≥12 consecutive months
 - Of these, 21 patients never received OAD treatment, and 12 patients discontinued OAD treatment for ≥12 consecutive months during SUSTAIN-3 (**Figure 2**)
 - Mean (SD) and median duration from study start to the start of the ESK monotherapy period was 242.9 (329.14) days and 84.0 days, respectively (range: 0-1,167 days)
- For the monotherapy group, the mean (SD) and median duration of exposure to ESK was 1,241 (550) days and 1,221 days, respectively, with a maximum duration of ESK exposure of 2,149 days (almost 6 years)
 - Mean (SD) and median duration of ESK monotherapy treatment was 926.2 (434.45) days and 875.0 days, respectively (range: 366-1,924 days)
- Baseline characteristics are shown in **Table 1**

Clinician- and patient-reported disease severity

- Changes in MADRS and PHQ-9 scores throughout the study period are shown in **Figure 3** and demonstrate that measures of depression severity remained stable throughout OP/M for patients receiving ESK monotherapy
- Mean changes in MADRS and PHQ-9 total scores from OP/M baseline to study end were similar between patients receiving ESK monotherapy and the overall population,[4] with mean (SD) changes in MADRS and PHQ-9 scores from OP/M baseline to endpoint of 0.5 (5.88) and 1.5 (4.72), respectively, in patients receiving ESK monotherapy, comparable to 0.2 (9.93) and 0.6 (6.22) in the overall SUSTAIN-3 population

Remission

- The proportion of patients who had received ESK monotherapy who achieved remission remained stable throughout the OP/M phase (**Figure 4**)
- By study end, 57.6% of patients who had received ESK monotherapy were in remission based on MADRS total score, compared with 49.6% of the overall SUSTAIN-3 population
- For remission based on PHQ-9, 42.4% of patients who had received ESK monotherapy were in remission by study end, compared with 33.9% of the overall SUSTAIN-3 population

Safety

- Overall, 72.7% (n = 24) of patients who had received ESK monotherapy experienced TEAEs possibly related to study drug, and 15.2% (n = 5) experienced ≥1 serious TEAE
- The most common TEAEs (≥20% of patients) are shown in **Table 2**
- Results from this subgroup analysis were consistent with the established safety and tolerability profile of ESK, with no new safety signals identified[6]

Figure 2: Flow chart of ESK monotherapy patients

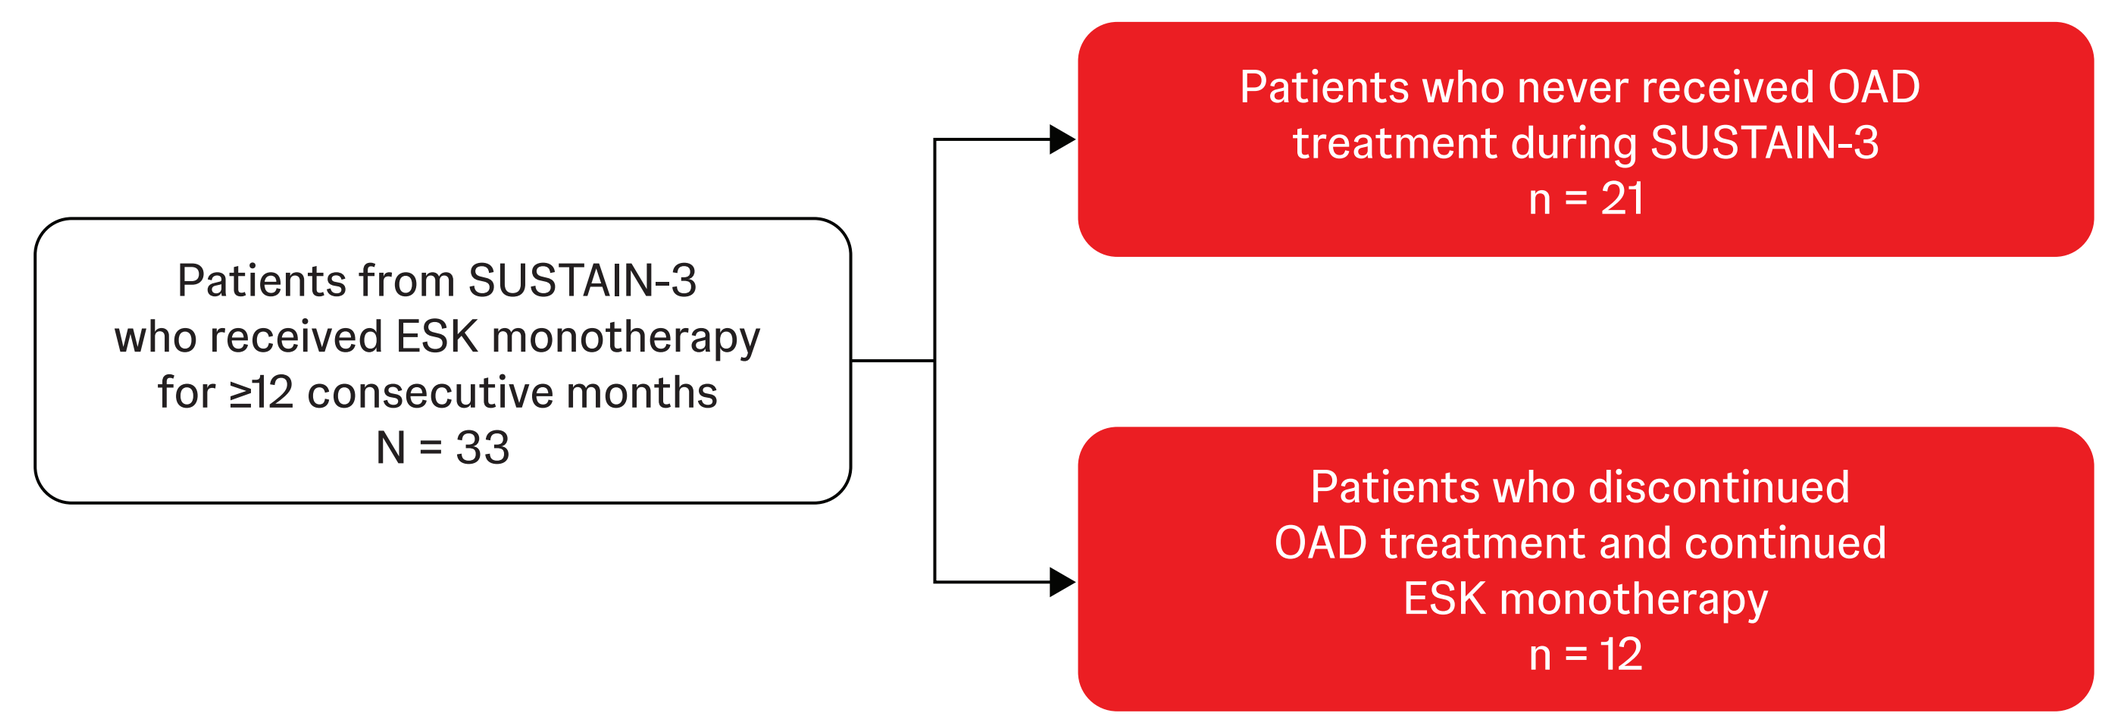
Patients from SUSTAIN-3 who received ESK monotherapy for ≥12 consecutive months N = 33
Patients who never received OAD treatment during SUSTAIN-3 n = 21
Patients who discontinued OAD treatment and continued ESK monotherapy n = 12

ESK, esketamine nasal spray; OAD, oral antidepressant.

Figure 3: Change in (A) MADRS and (B) PHQ-9 total scores over time for patients receiving ESK monotherapy (LOCF)

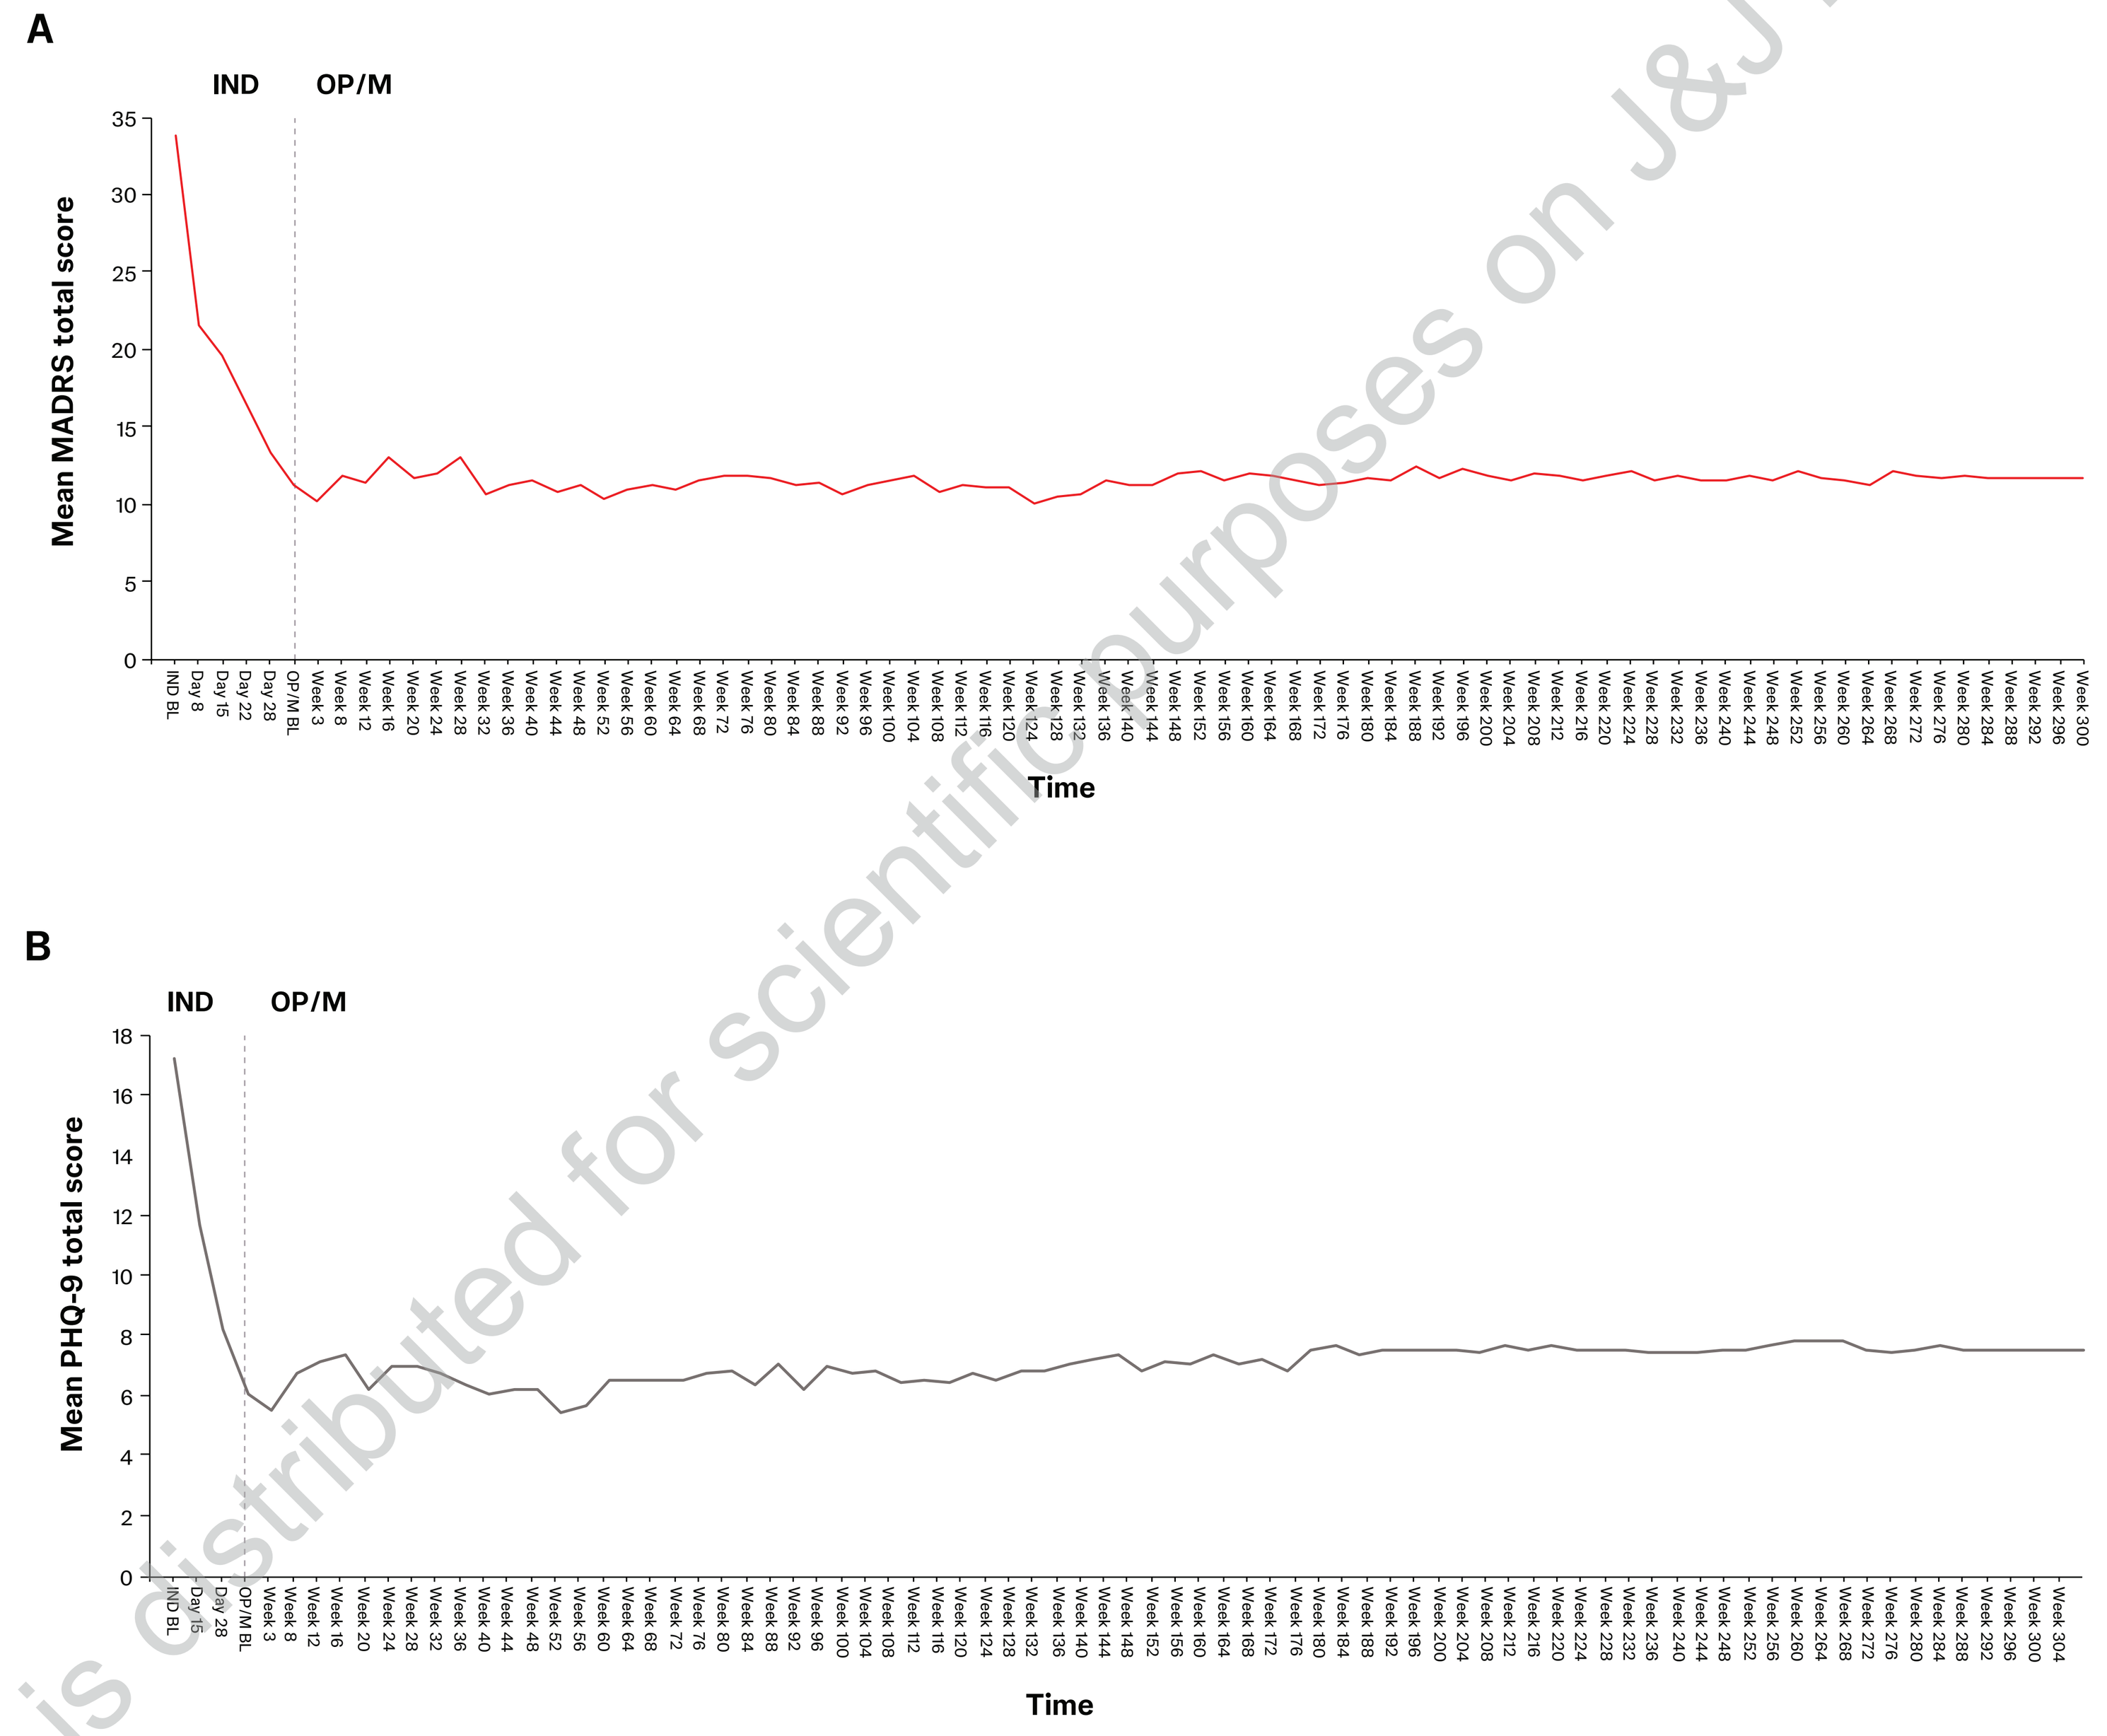
A
IND
OP/M
Mean MADRS total score
Time
B
IND
OP/M
Mean PHQ-9 total score
Time

BL, baseline; ESK, esketamine nasal spray; IND, induction phase; LOCF, last observation carried forward; MADRS, Montgomery-Åsberg Depression Rating Scale; OP/M, optimization/maintenance phase; PHQ-9, 9-item Patient Health Questionnaire.
Patients could enter SUSTAIN-3 at either the IND phase or OP/M phase, based on their status at the end of the parent study.

Figure 4: Proportion of patients receiving ESK monotherapy in remission based on (A) MADRS and (B) PHQ-9 total scores (LOCF)

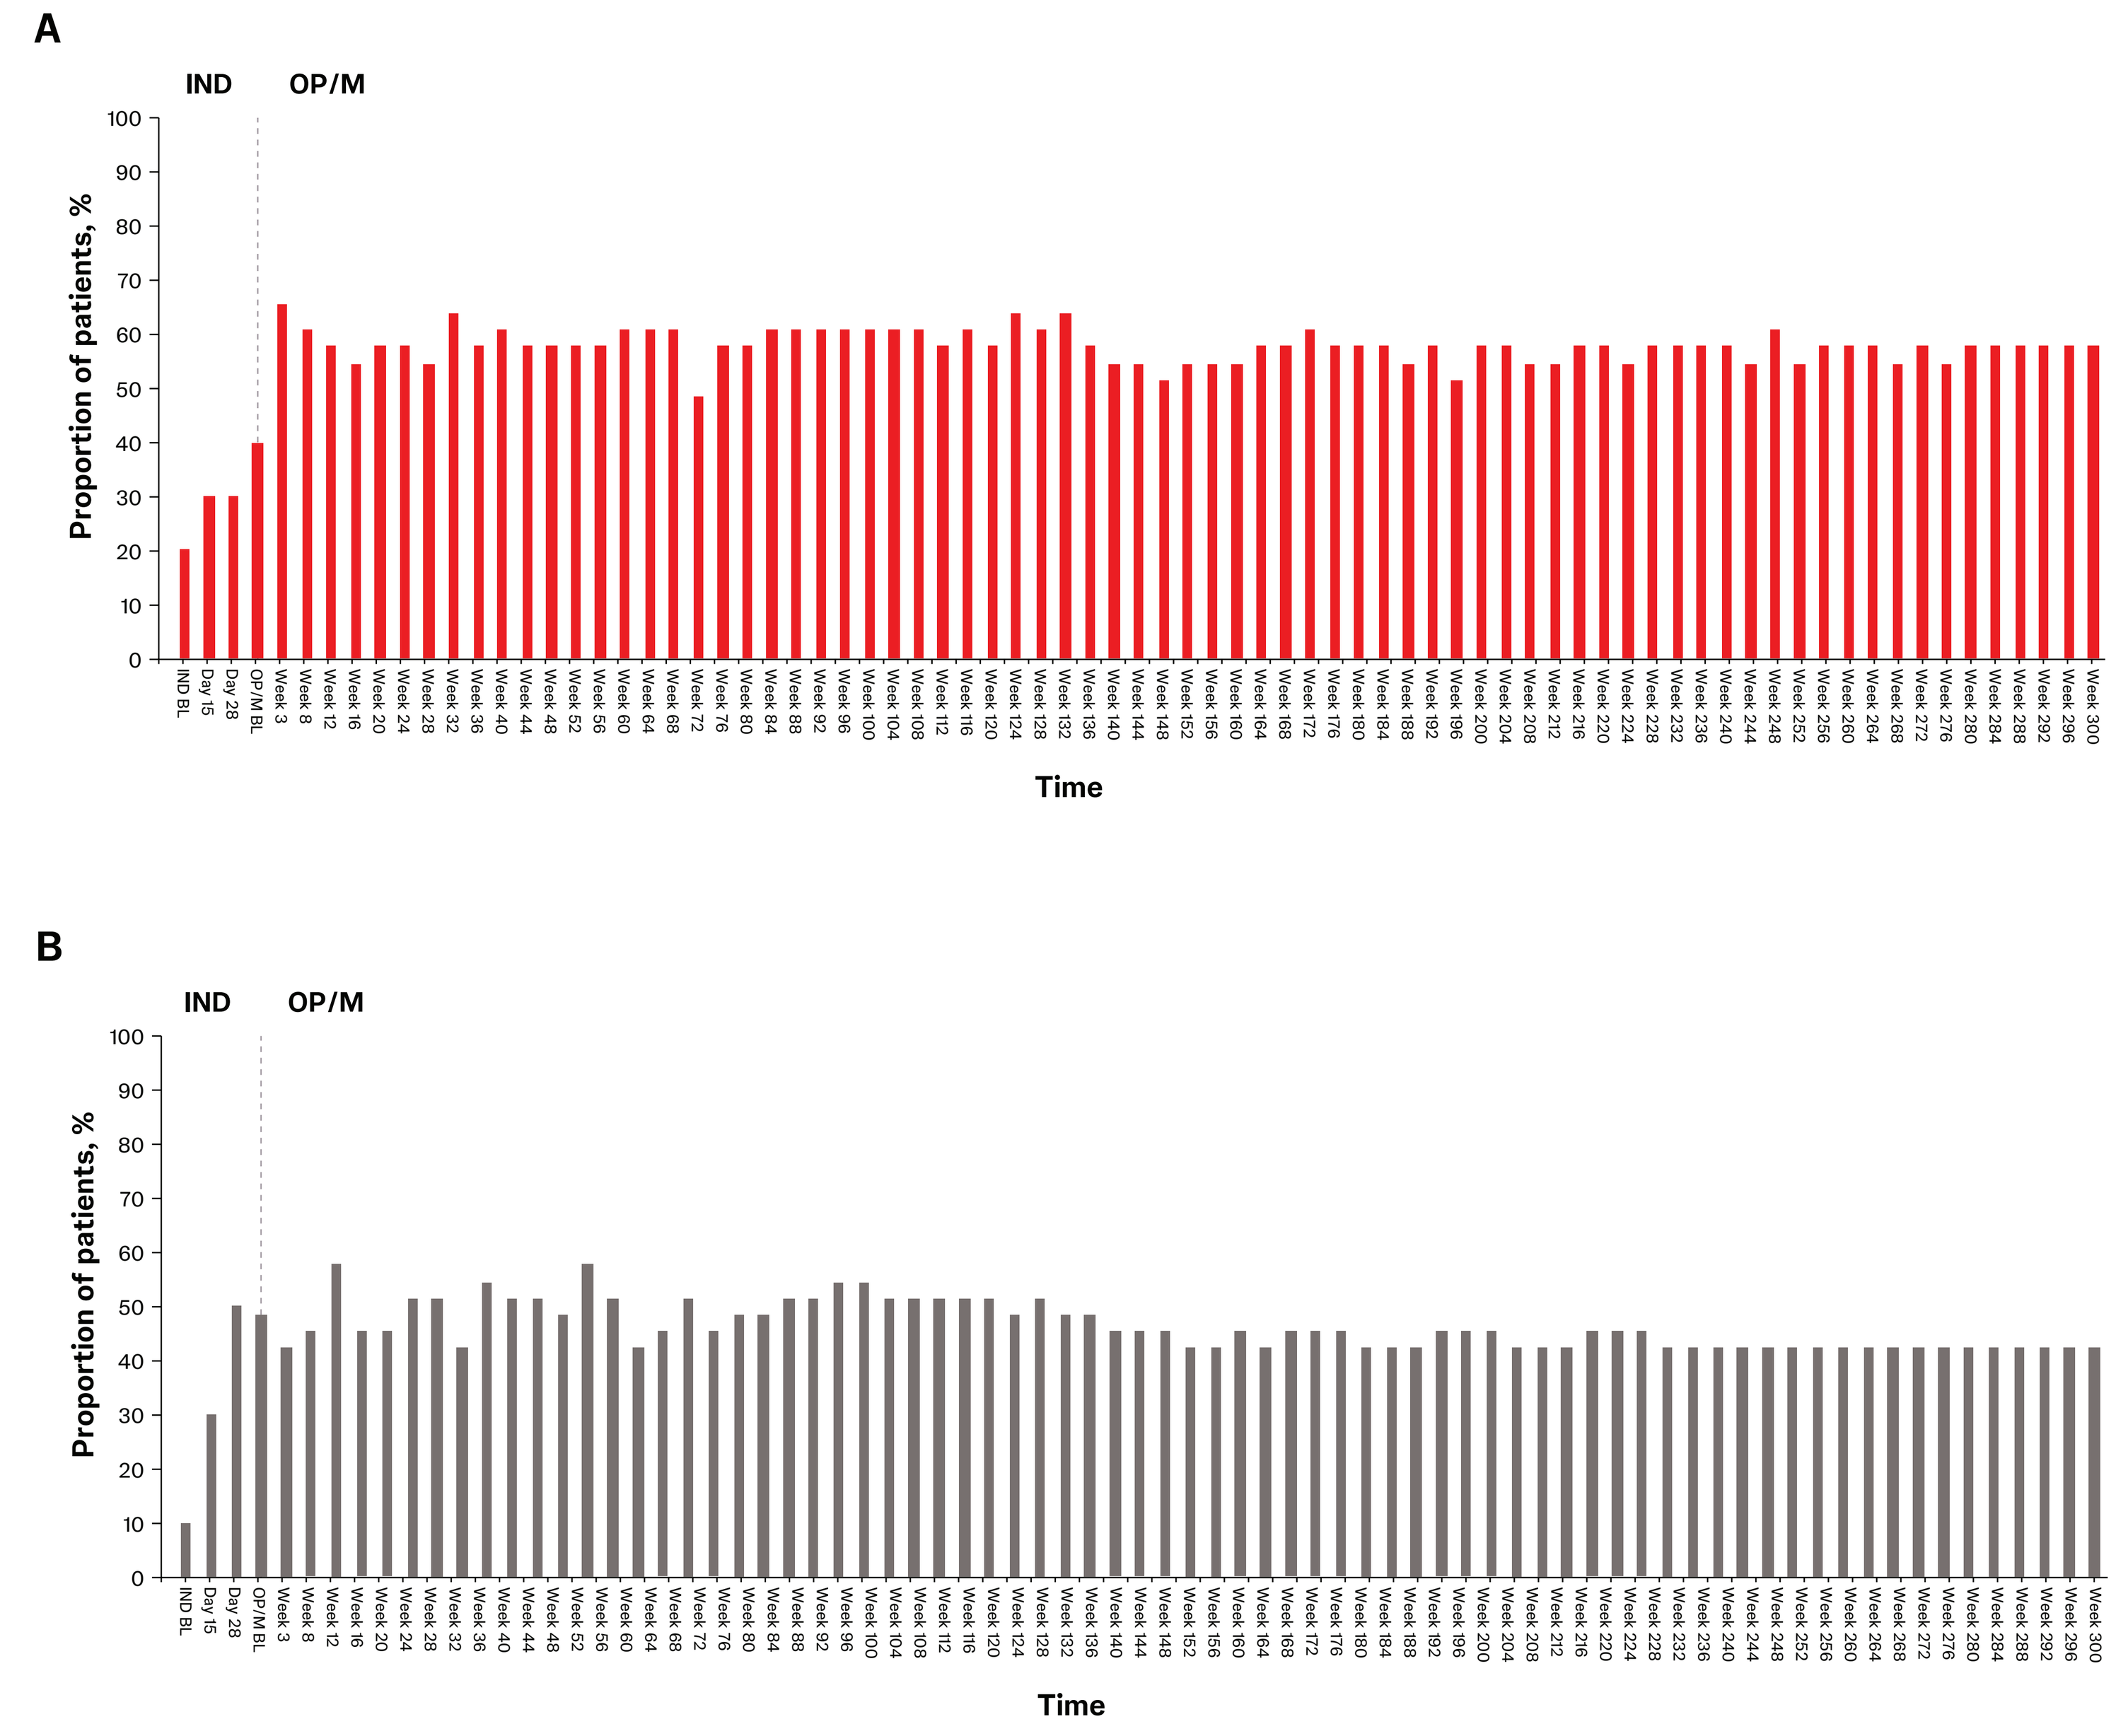
A
IND
OP/M
Proportion of patients, %
Time
B
IND
OP/M
Proportion of patients, %
Time

BL, baseline; ESK, esketamine nasal spray; IND, induction phase; LOCF, last observation carried forward; MADRS, Montgomery-Åsberg Depression Rating Scale; OP/M, optimization/maintenance phase; PHQ-9, 9-item Patient Health Questionnaire.
Patients could enter SUSTAIN-3 at either the IND phase or OP/M phase, based on their status at the end of the parent study.

Key Takeaway

Patients who received ESK as monotherapy without a concurrent OAD for ≥12 months had long-term stable depressive symptoms with no new safety signals identified

Conclusions

- Patients who received esketamine nasal spray monotherapy for ≥12 consecutive months at any point during the study remained stable during the optimization/maintenance phase
- Improvements in depressive symptoms were similarly maintained in patients receiving esketamine nasal spray monotherapy and the overall SUSTAIN-3 study population
- Esketamine nasal spray monotherapy had a tolerable safety profile, and no new safety signals were identified
- Esketamine nasal spray monotherapy may be a viable treatment option for those who receive little or no benefit from OADs, or who experience treatment-limiting TEAEs from OADs

Limitations

- SUSTAIN-3 is an open-label study with no control group for comparison
- This is a subgroup analysis with a small sample size, which may limit the interpretation of the results
- Patients' ESK treatment history differed depending on which parent study they participated in prior to SUSTAIN-3
- ESK monotherapy for a continuous time period was started at different stages of the study and was of variable duration (although ≥12 consecutive months)
- Patients had different reasons for initiating and continuing monotherapy treatment

Acknowledgments
The authors thank Laura van Laeren, PhD (ApotheCom, Yardley, PA), for editorial and writing assistance, which was funded by Johnson & Johnson.

Disclosures
JZ has received grants/research support from Boehringer Ingelheim, Compass, ElMindA, Cheryl T. Herman Foundation, F. Hoffman-La Roche, Jazz Pharmaceutical, Johnson & Johnson/Janssen, LivaNova, Otsuka, Neurocrine, Novartis, SAGE Therapeutics, and Takeda, and has served as a consultant or on advisory boards for Alpha Sigma, ElMindA, Johnson & Johnson/Janssen, Lundbeck, and Takeda. D-JF, IT, PC, OL, LL, MKF, and MA are employees of Janssen Scientific Affairs, LLC, a Johnson & Johnson company, and hold stock in Johnson & Johnson.

Previous presentation
Previously presented at Psych Congress Elevate, May 28-31, 2025 in Las Vegas, NV, USA.

Novel Pathways in Depression

Scan the QR code

The QR code is intended to provide scientific information for individual reference, and the information should not be altered or reproduced in any way.

This study was funded by Johnson & Johnson.

Presented at the Neuroscience Education Institute (NEI) Fall Congress, November 6-9, 2026; Colorado Springs, CO, USA

References
1. SPRAVATO® (esketamine) nasal spray, CIII. Prescribing information. Janssen Pharmaceuticals, Inc; 2025. Accessed September 2, 2025. https://www.janssenlabels.com/package-insert/product-monograph/prescribing-information/SPRAVATO-pi.pdf 2. Janik A et al. *JAMA* Psychiatry. 2025 Jul 2:e251317. doi: 10.1001/jamapsychiatry. 2025.1317 3. SPRAVATO® (esketamine) approved in the U.S. as the first and only monotherapy for adults with treatment-resistant depression. News release. Johnson & Johnson; 2025. Accessed September 2, 2025. https://www.jnj.com/media-center/press-releases/spravato-esketamine-approved-in-the-u-s-as-the-first-and-only-monotherapy-for-adults-with-treatment-resistant-depression 4. Zaki N et al. *Neuropsychopharmacology.* 2023;48:1225-1233. 5. Zaki N et al. *Int J Neuropsychopharmacol*. 2025; pyaf027. Accessed September 2, 2025. https://doi.org/10.1093/ijnp/pyaf027. 6. McIntyre RS et al. *Eur Neuropsychopharmacol*. 2024;85:58-65.

This material is distributed for scientific purposes on J&J Medical Connect and is not for promotional use