Change in Clinician-Rated and Patient-Reported Depression Severity After Esketamine Nasal Spray as Monotherapy in Adult Patients With Treatment-Resistant Depression: A Post Hoc Analysis of Item Scores

Richard Shelton[1], Mai Himedan[2], Dong-Jing Fu[2], Ibrahim Turkoz[2], Amit Patel[2], Lisa Lim[2], Oliver Lopena[2]

[1]University of Alabama at Birmingham, Birmingham, AL; [2]Johnson & Johnson, Titusville, NJ

This material is distributed for scientific purposes on J&J Medical Connect, and is not for promotional use

Introduction

- Morbidity and mortality associated with depression is disproportionately higher for patients with treatment-resistant depression (TRD), commonly defined as inadequate response to at least 2 antidepressants of adequate dose, duration, and adherence to treatment[1]
 - For many patients with TRD, depressive symptoms remain burdensome despite treatment with existing antidepressant medications[1]
- Esketamine nasal spray (ESK) was initially approved, in conjunction with an oral antidepressant (OAD), in 2019 for the treatment of TRD in adults and in 2020 for the treatment of depressive symptoms in adults with major depressive disorder with acute suicidal ideation or behavior[2]
- Primary findings from a phase 4, multicenter, double-blind, randomized, placebo-controlled study (NCT04599855) demonstrated that ESK as monotherapy led to a rapid and superior improvement in Montgomery-Åsberg Depression Rating Scale (MADRS) total score compared with placebo at day 28[3]
 - Based on results of this study, ESK was approved in the United States as the first and only monotherapy for adults with TRD in January 2025[2,4]

Objective

- The objective of this post hoc analysis was to evaluate changes in items of the MADRS and 9-item Patient Health Questionnaire (PHQ-9) over 4 weeks of ESK monotherapy

Methods

Study design

- This multicenter, double-blind, randomized placebo-controlled phase 4 study (NCT04599855) included adults (≥18 years) with major depressive disorder and a history of inadequate response (≤25% improvement) to ≥2 different OADs
 - Inadequate response to OADs was assessed using the Massachusetts General Hospital Antidepressant Treatment Response Questionnaire
 - Potential participants were excluded from enrollment if they had suicidal behavior in the last year, or suicidal ideation with some intent to act within 6 months prior to screening
- The study had 4 phases: screening, double-blind treatment, open-label treatment/observation, and follow-up (**Figure 1**)
 - During the 4-week double-blind phase, patients were randomly assigned to receive monotherapy with fixed doses of ESK 56 mg, ESK 84 mg, or placebo nasal spray (PBO) twice weekly

Figure 1: Study design

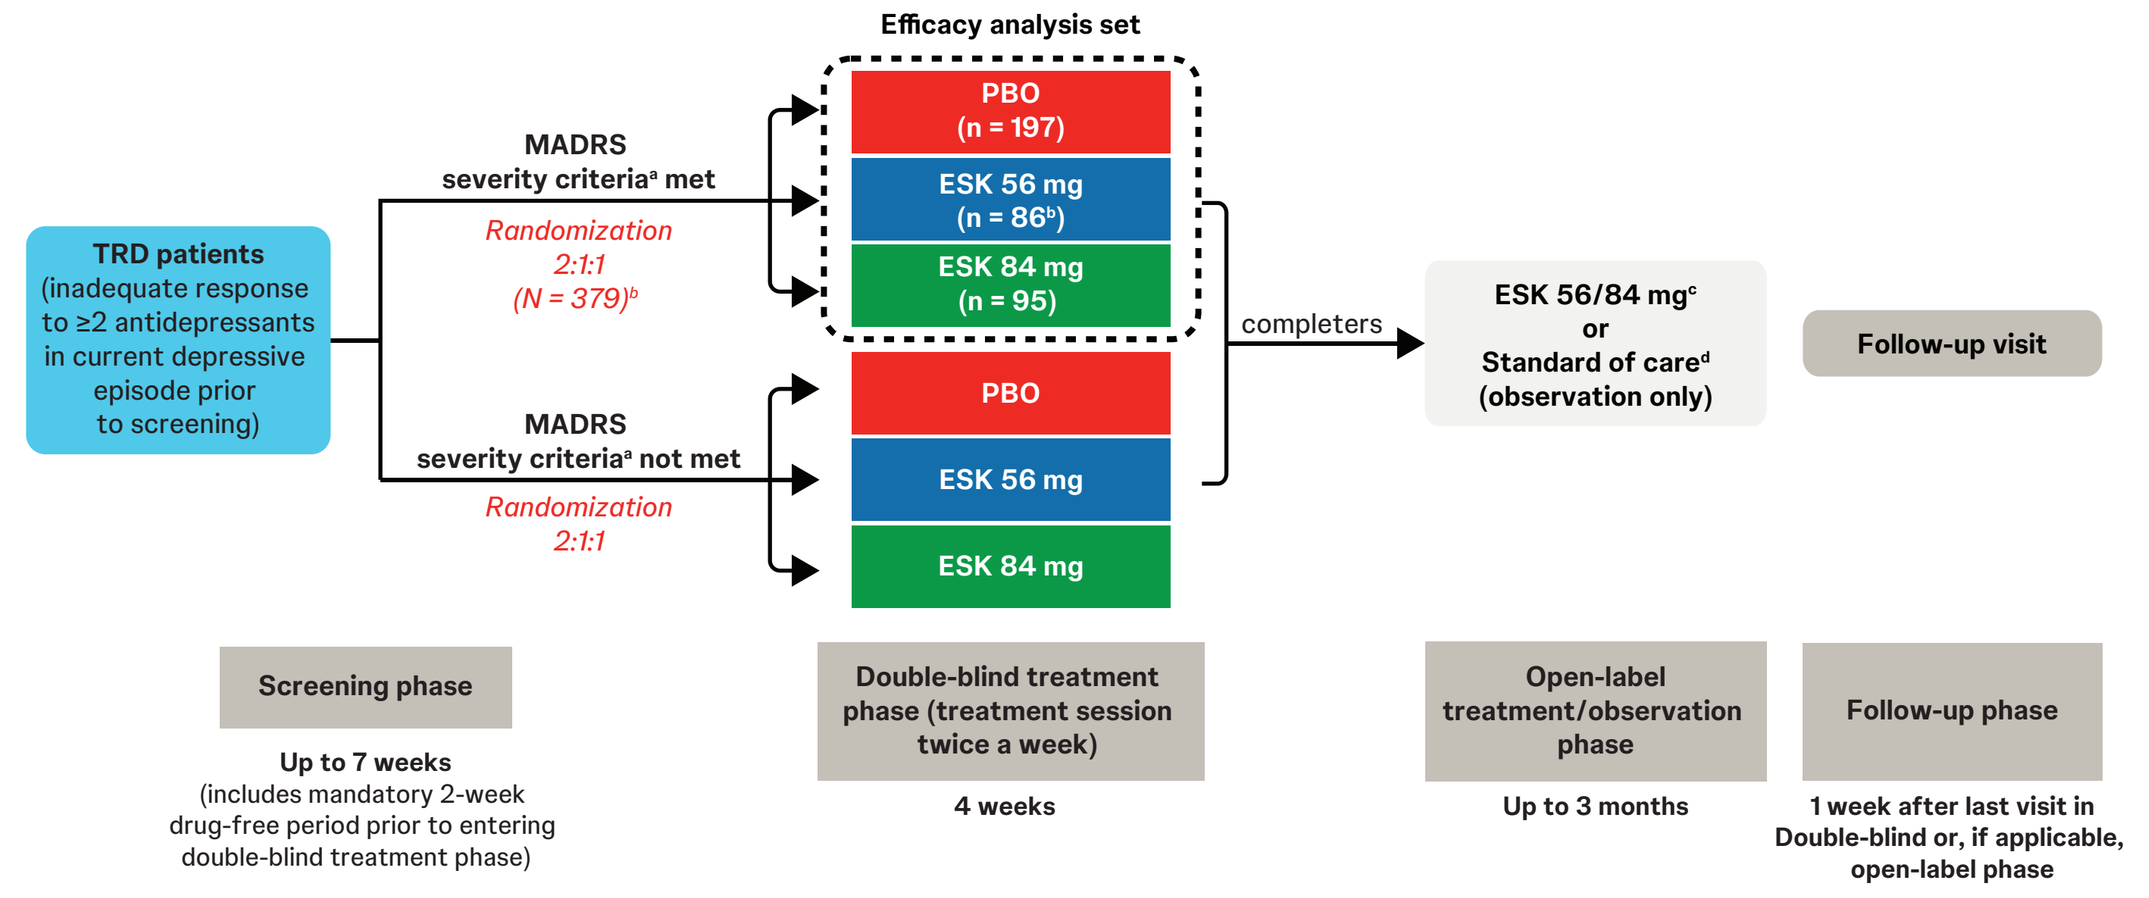
Efficacy analysis set
TRD patients (inadequate response to ≥2 antidepressants in current depressive episode prior to screening)
MADRS severity criteria[a] met
Randomization 2:1:1 (N = 379)[b]
MADRS severity criteria[a] not met
Randomization 2:1:1
PBO (n = 197)
ESK 56 mg (n = 86[b])
ESK 84 mg (n = 95)
PBO
ESK 56 mg
ESK 84 mg
completers
ESK 56/84 mg[c] or Standard of care[d] (observation only)
Follow-up visit
Screening phase
Up to 7 weeks (includes mandatory 2-week drug-free period prior to entering double-blind treatment phase)
Double-blind treatment phase (treatment session twice a week)
4 weeks
Open-label treatment/observation phase
Up to 3 months
Follow-up phase
1 week after last visit in Double-blind or, if applicable, open-label phase

ESK, esketamine nasal spray; MADRS, Montgomery-Åsberg Depression Rating Scale; PBO, placebo nasal spray; TRD, treatment-resistant depression.
[a]MADRS total score ≥28 at screening week 1, week 2, and day 1 (pre-randomization) and ≤25% improvement in the MADRS total score from screening week 1 to day 1 (pre-randomization); referred to as "nonresponse criteria" in the protocol.
[b]One of 87 patients randomly assigned to ESK 56 mg did not receive a dose of study drug and was not included in the efficacy analysis dataset.
[c]With or without standard-of-care treatment.
[d]Only 1 patient received standard-of-care treatment without ESK.
Current antidepressant medications (including adjunctive treatments) were tapered during the screening phase, resulting in a ≥2-week antidepressant (and antipsychotic)-free observation period immediately prior to randomization.

Assessments

- Baseline floor effects (score = 0) for the MADRS and PHQ-9 items were evaluated using item-level frequency distributions
- Least squares (LS) mean change from baseline in MADRS and PHQ-9 items between treatment groups were evaluated using analysis of covariance models with change from baseline as the dependent variable, treatment as factor, and screening antidepressant status and baseline value as covariates
 - Individual MADRS item scores were evaluated on days 2, 15, and 28
 - Individual PHQ-9 item scores were evaluated on days 15 and 28
- Based on item score distributions, item responders (those with at least a 1-point improvement) were examined using logistic regression
 - Logistic regression, odds ratios and corresponding 95% confidence intervals were provided
- Treatment-emergent adverse events (TEAEs) were monitored for the duration of the study

Results

Baseline characteristics

- The analysis included 378 participants, of which 197 received PBO, 86 received ESK 56 mg and 95 received ESK 84 mg
- Demographic and baseline characteristics were comparable between the treatment arms (**Table 1**)
 - At baseline, most patients were female (61.1%), with a mean age of 45.4 years and mean baseline MADRS and PHQ-9 total scores of 37.3 and 20.0, respectively
 - Mean duration of current depressive episode was numerically lower in the PBO group (289 days) compared with the ESK groups (406-419 days)
- Mean MADRS and PHQ-9 items scores at baseline are shown in **Table 2**

Table 1: Baseline demographic and disease characteristics

Baseline characteristic	PBO n = 197	ESK 56 mg n = 86	ESK 84 mg n = 95	Total N = 378
Mean age (SD), years	45.2 (13.8)	46.5 (14.2)	44.8 (14.7)	45.4 (14.1)
Female, n (%)	119 (60.4)	51 (59.3)	61 (64.2)	231 (61.1)
Race, n (%)				
American Indian or Alaska Native	0	1 (1.2)	0	1 (0.3)
Asian	5 (2.5)	2 (2.3)	4 (4.2)	11 (2.9)
Black or African American	13 (6.6)	4 (4.7)	8 (8.4)	25 (6.6)
Native Hawaiian/Pacific Islander	0	1 (1.2)	1 (1.1)	2 (0.5)
White	171 (86.8)	76 (88.4)	81 (85.3)	328 (86.8)
Multiple[a]	4 (2.0)	1 (1.2)	1 (1.1)	6 (1.6)
Not reported/unknown	4 (2.0)	1 (1.2)	0	5 (1.4)
Mean duration of current episode (range), weeks	289.0 (10-1872)	419.8 (12-2555)	406.4 (10-2236)	348.3 (10-2555)
Number of failed antidepressant interventions,[b] n (%)				
2	117 (59.4)	49 (57.0)	58 (61.1)	224 (59.3)
≥3	80 (40.6)	37 (43.0)	37 (38.9)	154 (40.7)
Mean baseline MADRS total score (SD)	37.5 (4.90)	37.5 (5.23)	36.6 (4.48)	37.3 (4.88)
Mean baseline PHQ-9 total score (SD)	19.8 (4.07)	20.7 (3.43)	19.9 (3.79)	20.0 (3.87)

ESK, esketamine nasal spray; MADRS, Montgomery-Åsberg Depression Rating Scale; PBO, placebo nasal spray; PHQ-9, 9-item Patient Health Questionnaire.
[a]If multiple race categories were selected, the race was recorded as "Multiple."
[b]Failed antidepressant intervention history (defined as ≤25% improvement) taken for at least 6 weeks during the current episode as obtained in the Massachusetts General Hospital Antidepressant Treatment Response Questionnaire.

Efficacy

- All MADRS item scores improved from baseline to day 28 with ESK versus PBO (**Figure 2**)
 - MADRS items of reported sadness and apparent sadness were significantly improved in both ESK groups compared with PBO on all days ($P < 0.05$)
 - By day 28, most MADRS items were significantly improved in both ESK dose groups compared with PBO ($P < 0.05$), including reported sadness, apparent sadness, inner tension, concentration difficulties, lassitude, and inability to feel
 - The item of suicidal thoughts showed significant improvement with ESK 84 mg by day 28
 - Several MADRS items had significant ($P < 0.05$) and rapid improvement by day 2, including reported sadness, apparent sadness, inner tension, and pessimistic thoughts (both ESK groups), as well as lassitude and inability to feel (ESK 84 mg)
- The likelihood of achieving a ≥1-point improvement from baseline for MADRS item scores was assessed at days 2 and 28, and favored ESK over PBO for all items except reduced appetite at day 2 (**Figure 3**)
 - For most items, there was a greater likelihood of achieving a ≥1-point improvement from baseline in the ESK 84-mg group compared with the ESK 56-mg group
 - At both days 2 and 28, the likelihood of achieving a ≥1-point improvement was significant for items including reported sadness, apparent sadness, lassitude, concentration difficulties, and pessimistic thoughts
 - At day 28, the likelihood of achieving a ≥1-point improvement was significant for additional items, including inability to feel, reduced sleep, reduced appetite, and suicidal thoughts
- All PHQ-9 items scores improved from baseline to day 28 with ESK versus placebo (**Table 3**)
 - On day 28, items of little interest or pleasure in doing things, feeling down or depressed or hopeless, trouble falling or staying asleep, feeling tired or little energy, feeling bad about yourself, trouble concentrating on thing, and moving slowly or fidgety or restlesss were significantly decreased on day 28 in both ESK groups (all $P < 0.05$ vs PBO)

Safety

- The most commonly reported TEAEs were nausea, dissociation, dizziness, and headache[2]
- Serious TEAEs were reported in 6 patients in the double-blind phase: ESK 56 mg: ankle fracture (n = 1); ESK 84 mg: ophthalmic migraine and suicide attempt (n = 1 each); PBO: self-injurious ideation, suicidal ideation, and acute myocardial infarction (n = 1 each). None of these (except acute myocardial infarction: PBO), were considered related to the study medication[2]
- No deaths were reported in either the double-blind or open-label phase[2]

Figure 2: LS mean change (±SE) from baseline in MADRS item scores over time (excluding patients with a baseline score of 0)

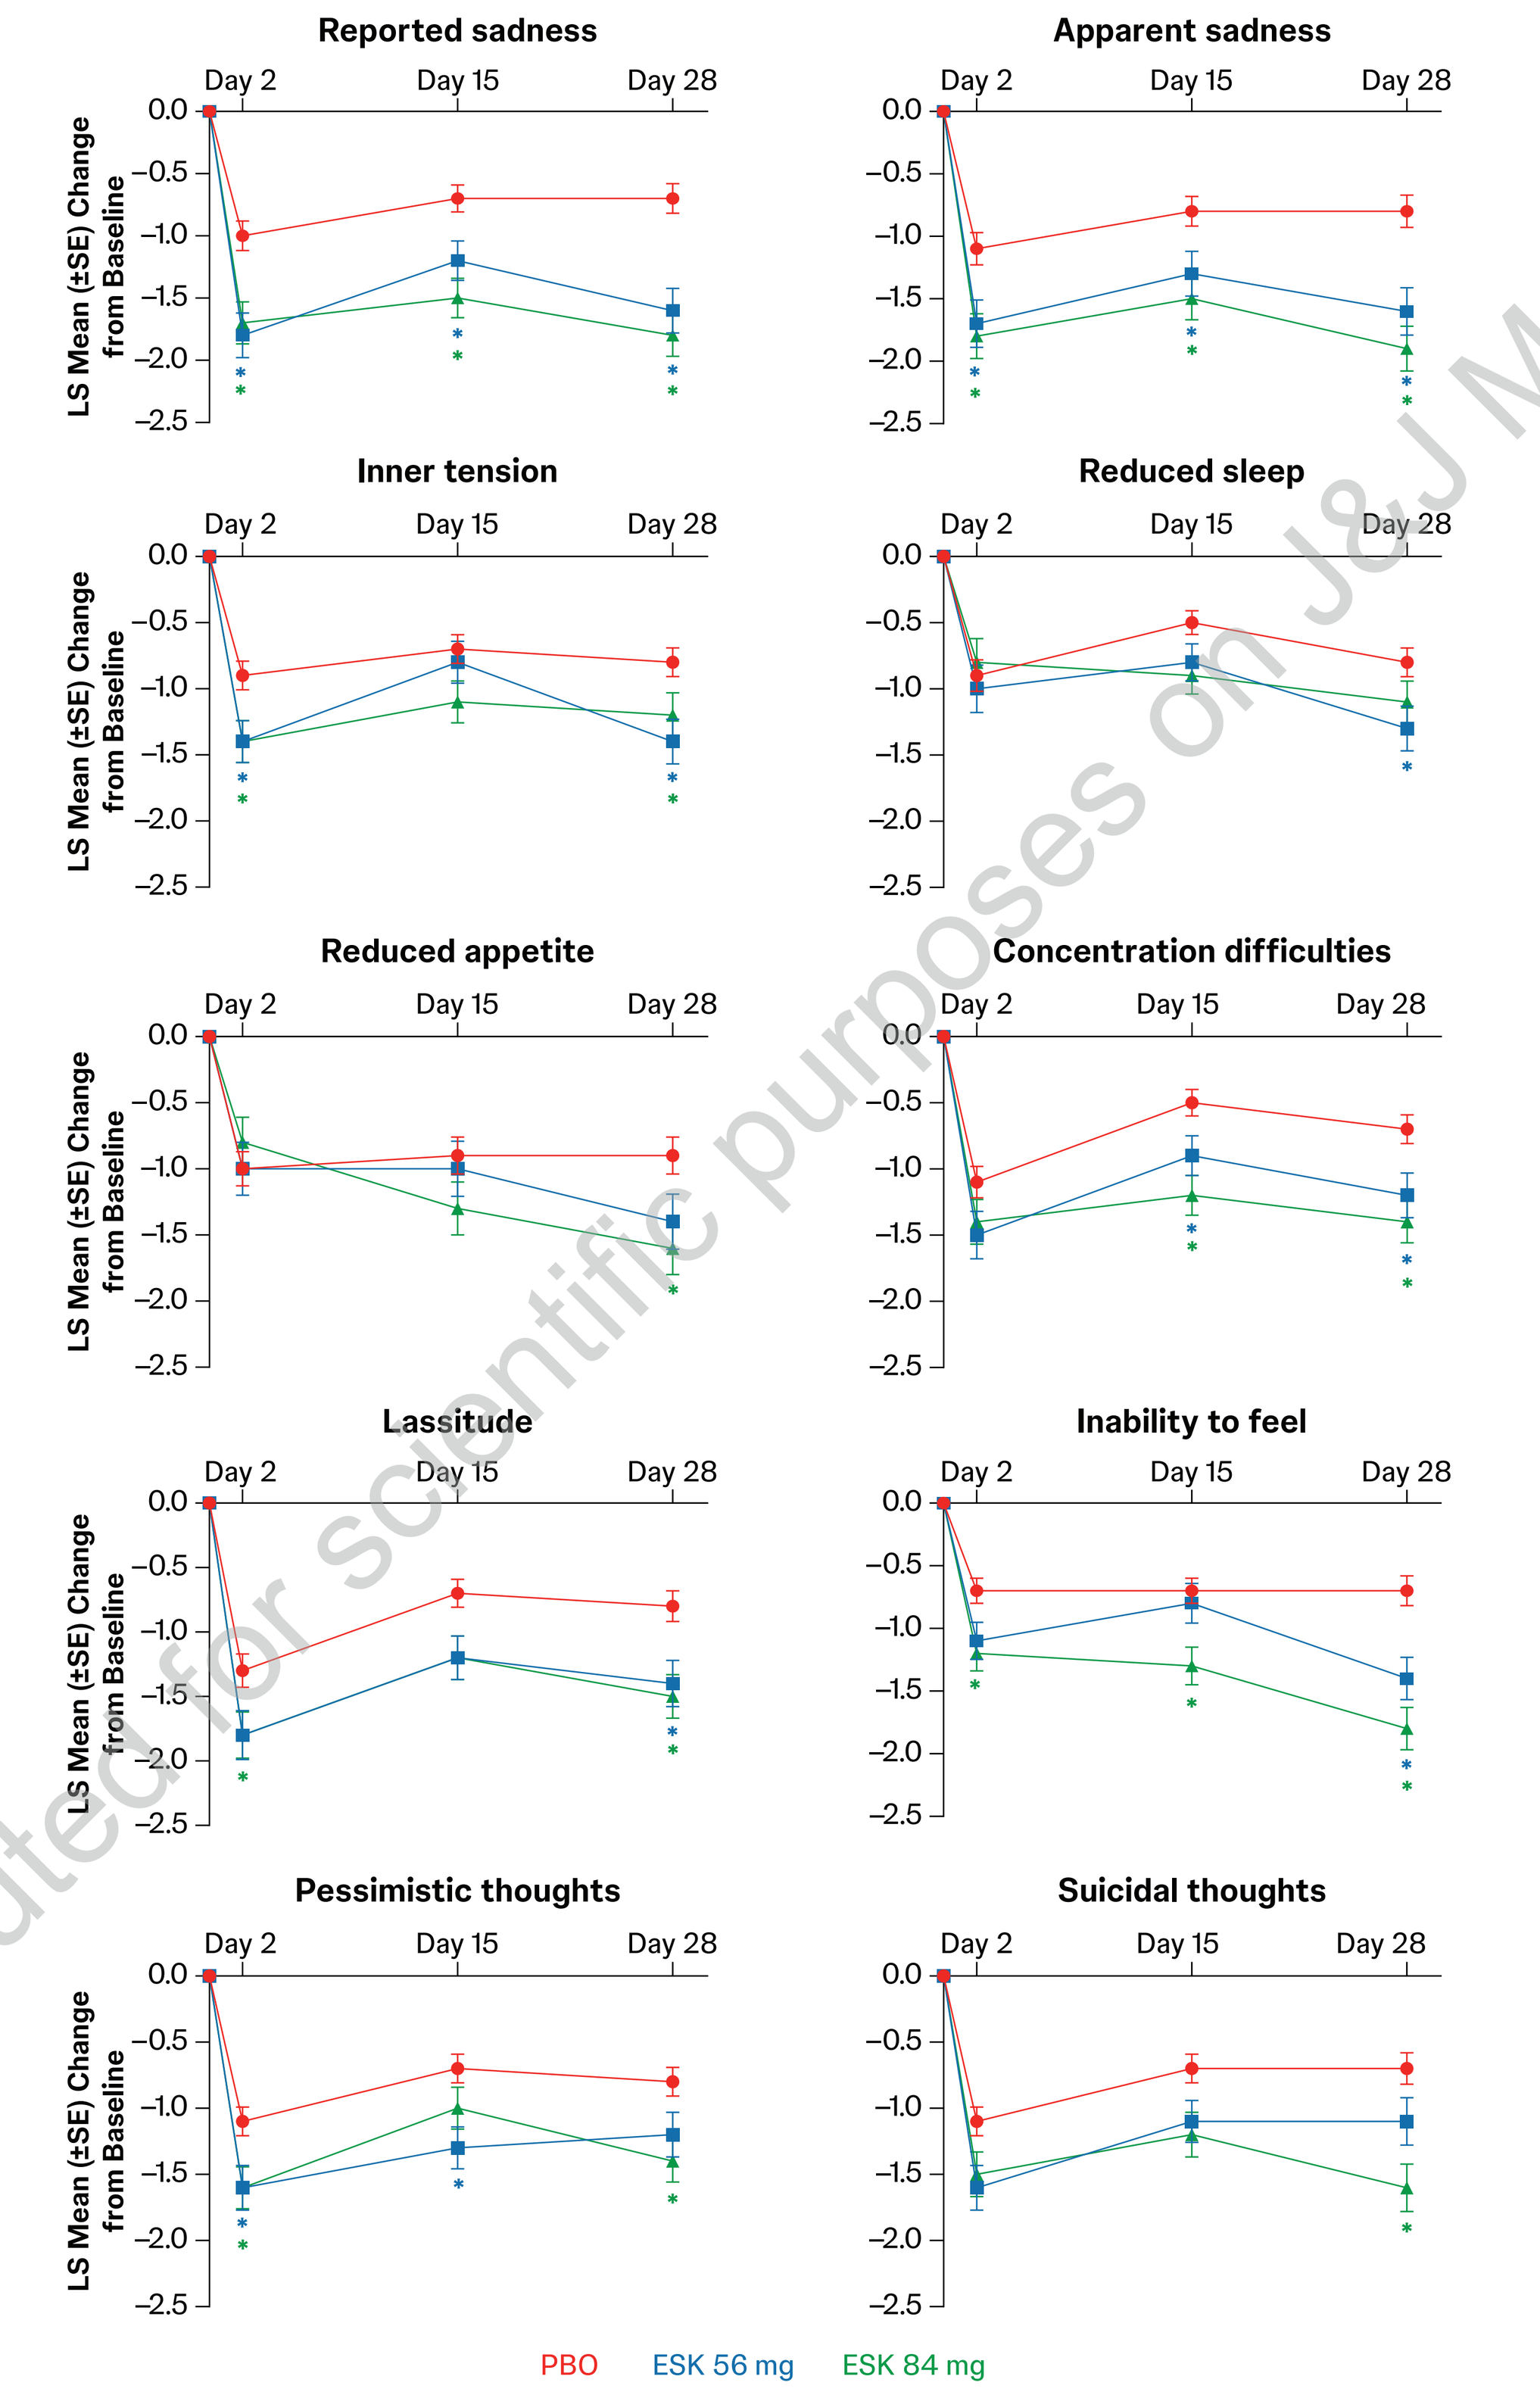
Reported sadness
Apparent sadness
Inner tension
Reduced sleep
Reduced appetite
Concentration difficulties
Lassitude
Inability to feel
Pessimistic thoughts
Suicidal thoughts
Day 2
Day 15
Day 28
LS Mean (±SE) Change from Baseline
0.0
−0.5
−1.0
−1.5
−2.0
−2.5
PBO
ESK 56 mg
ESK 84 mg

ESK, esketamine nasal spray; LS, least squares; MADRS, Montgomery-Åsberg Depression Rating Scale; PBO, placebo nasal spray.
*$P < 0.05$ vs PBO.

Table 2: Baseline MADRS and PHQ-9 item scores

	PBO n = 197	ESK 56 mg n = 86	ESK 84 mg n = 95
MADRS baseline item score / proportion of patients with a baseline score of 0, mean (SD) / n (%)			
Reported sadness	4.6 (0.72) / 0	4.8 (0.66) / 0	4.7 (0.71) / 0
Apparent sadness	4.3 (1.0) / 3 (1.5)	4.4 (0.83) / 0	4.3 (0.80) / 0
Inner tension	3.8 (1.14) / 5 (2.5)	3.6 (1.34) / 5 (5.8)	3.5 (1.40) / 9 (9.5)
Reduced sleep	3.8 (1.23) / 4 (2.0)	3.9 (1.24) / 2 (2.3)	3.7 (1.43) / 7 (7.4)
Reduced appetite	2.9 (1.86) / 40 (20.3)	2.9 (1.95) / 20 (23.3)	2.5 (1.71) / 22 (23.2)
Concentration difficulties	4.0 (1.27) / 10 (5.1)	4.1 (1.24) / 4 (4.7)	3.9 (1.02) / 3 (3.2)
Lassitude	4.2 (1.00) / 2 (1.0)	4.2 (0.97) / 1 (1.2)	4.3 (0.78) / 0
Inability to feel	4.3 (0.72) / 0	4.1 (0.82) / 0	4.2 (0.83) / 0
Pessimistic thoughts	3.8 (0.95) / 4 (2.0)	3.7 (1.03) / 2 (2.3)	3.6 (1.08) / 2 (2.1)
Suicidal thoughts	1.8 (1.48) / 58 (29.4)	2.0 (1.66) / 27 (31.4)	1.8 (1.59) / 34 (35.8)
PHQ-9 baseline item score / proportion of patients with a baseline score of 0, mean (SD) / n (%)			
Little interest or pleasure in doing things	2.5 (0.70) / 2 (1.0)	2.7 (0.54) / 1 (1.2)	2.6 (0.64) / 1 (1.1)
Feeling down or depressed or hopeless	2.7 (0.60) / 1 (0.5)	2.8 (0.49) / 0	2.8 (0.44) / 0
Trouble falling or staying asleep	2.5 (0.75) / 4 (2.0)	2.7 (0.65) / 1 (1.2)	2.6 (0.69) / 1 (1.1)
Feeling tired or little energy	2.7 (0.60) / 1 (0.5)	2.7 (0.55) / 0	2.7 (0.64) / 2 (2.1)
Poor appetite or overeating	2.1 (1.02) / 21 (10.7)	2.4 (0.86) / 5 (5.8)	2.0 (1.08) / 12 (12.6)
Feeling bad about yourself	2.5 (0.79) / 4 (2.0)	2.5 (0.84) / 3 (3.5)	2.4 (0.88) / 5 (5.3)
Trouble concentrating on things	2.5 (0.77) / 4 (2.0)	2.4 (0.90) / 6 (7.0)	2.4 (0.87) / 4 (4.2)
Moving slowly or fidgety or restless	1.5 (1.10) / 50 (25.4)	1.5 (1.15) / 23 (26.7)	1.3 (1.05) / 26 (27.4)
Thoughts you'd be better off dead	0.9 (1.1) / 106 (53.8)	1.0 (1.15) / 43 (50.0)	1.0 (1.13) / 45 (47.4)

ESK, esketamine nasal spray; MADRS, Montgomery-Åsberg Depression Rating Scale; PBO, placebo nasal spray; PHQ-9, 9-item Patient Health Questionnaire.

Figure 3: Likelihood of achieving ≥1-point improvement in MADRS item score from baseline at days 2 and 28 (excluding patients with a baseline score of 0)

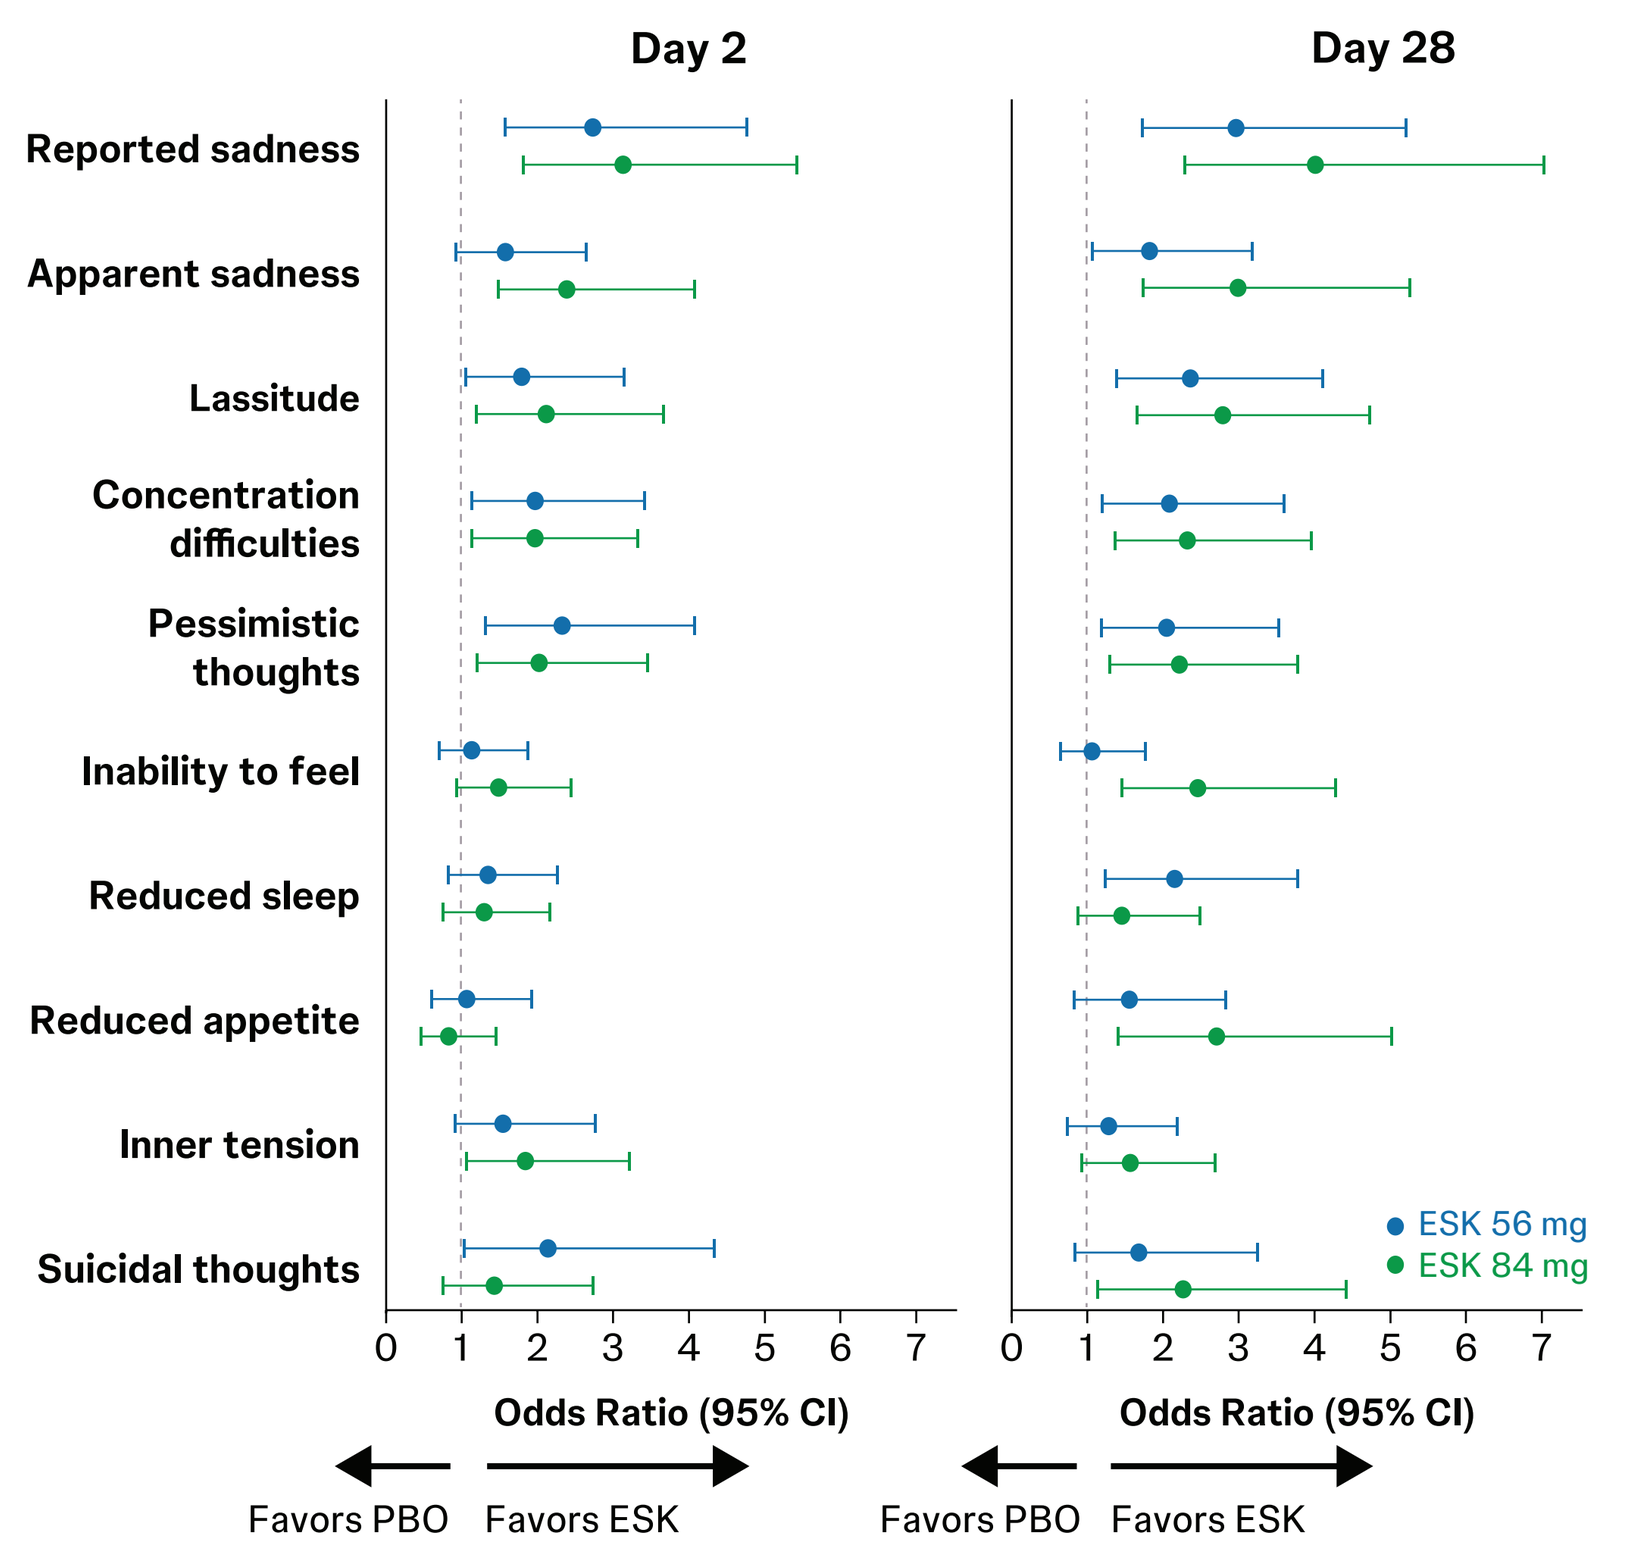
Day 2
Day 28
Reported sadness
Apparent sadness
Lassitude
Concentration difficulties
Pessimistic thoughts
Inability to feel
Reduced sleep
Reduced appetite
Inner tension
Suicidal thoughts
0 1 2 3 4 5 6 7
Odds Ratio (95% CI)
Favors PBO
Favors ESK
ESK 56 mg
ESK 84 mg

ESK, esketamine nasal spray; MADRS, Montgomery-Åsberg Depression Rating Scale; PBO, placebo nasal spray.
Unadjusted odds ratio for ≥1-point improvement in MADRS item score from baseline for ESK treatment (56 or 84 mg) compared with PBO.

Table 3: LS mean change (±SE) from baseline in PHQ-9 item scores at Day 28 (excluding patients with a baseline score of 0)

PHQ-9 item, LS mean (±SE) change from baseline	PBO n = 197	ESK 56 mg n = 86	ESK 84 mg n = 96
Little interest or pleasure in doing things	-0.3 (0.07)	-0.8 (0.10)*	-1.0 (0.10)*
Feeling down or depressed or hopeless	-0.5 (0.07)	-1.0 (0.11)*	-1.1 (0.10)*
Trouble falling or staying asleep	-0.3 (0.07)	-0.7 (0.11)*	-0.8 (0.11)*
Feeling tired or little energy	-0.3 (0.07)	-0.7 (0.10)*	-0.7 (0.10)*
Poor appetite or overeating	-0.4 (0.08)	-1.0 (0.12)*	-0.6 (0.12)
Feeling bad about yourself	-0.4 (0.07)	-1.0 (0.11)*	-0.9 (0.11)*
Trouble concentrating on things	-0.4 (0.07)	-0.8 (0.11)*	-0.9 (0.10)*
Moving slowly or fidgety or restless	-0.4 (0.08)	-1.0 (0.13)*	-1.0 (0.13)*
Thoughts you'd be better off dead	-0.5 (0.11)	-0.6 (0.15)	-1.2 (0.15)*

ESK, esketamine nasal spray; LS, least squares; PBO, placebo nasal spray; PHQ-9, 9-item Patient Health Questionnaire.
*$P < 0.05$ vs PBO.

Key Takeaway

ESK monotherapy improved depression severity as early as day 2 in all items of MADRS and continued through day 28

Conclusions

- All MADRS and PHQ-9 items improved from baseline to day 28 with esketamine nasal spray
- Both dose levels of esketamine nasal spray led to numerically greater improvement in all MADRS and PHQ-9 items compared with PBO
- No new safety signals were identified

Limitations

- This was a post hoc analysis of a randomized controlled trial that was not designed to assess changes in individual MADRS and PHQ-9 items and not powered for significance
- PHQ-9 results should be interpreted with caution, as evaluation of individual items has not been validated
- Some items had low symptomatology due to the nature of illness (sleep or appetite) or exclusion criteria of the study (acute suicidality)

Acknowledgments
The authors thank Lynn Brown, PhD (ApotheCom, Yardley, PA), for editorial and writing assistance, which was funded by Johnson & Johnson.

Disclosures
RS has received grants/research support from National Institutes of Mental Health, Patient-Centered Outcomes Research Institute, AbbVie Inc., Alto Pharmaceuticals, Boehringer Ingelheim, Denovo Biopharma, InMune Bio, Intra-cellular Therapies, Johnson & Johnson Innovative Medicine, LivaNova PLC, Navitor Pharmaceuticals, Neumora, Neurocrine Biosciences, Neurorx, Novartis AG, Otsuka Pharmaceuticals, Sumitomo Group, and Supernus Pharmaceuticals; has been a consultant for Boehringer Ingelheim, Denovo Biopharma, Evecxia Therapeutics LLC, Johnson & Johnson Innovative Medicine, Neurorx, Novartis AG, Otsuka Pharmaceuticals, Seelos Therapeutics, Inc., Sumitomo Group, and Supernus Pharmaceuticals; and has received royalties from Springer-Nature Group and Wolters-Kluwer NV. MH, D-JF, IT, AP, LL, and OL are employees of Johnson & Johnson, and hold stock in Johnson & Johnson.

Previous presentation
Previously presented at Psych Congress Elevate, May 28-31, 2025 in Las Vegas, NV, USA.

Novel Pathways in Depression

Scan the QR code

The QR code is intended to provide scientific information for individual reference, and the information should not be altered or reproduced in any way.

This study was funded by Johnson & Johnson.

Presented at the Neuroscience Education Institute (NEI) Fall Congress, November 6-9, 2025; Colorado Springs, CO, USA

References
1. McIntyre RS et al. *World Psychiatry*. 2023;22(3):394-412. 2. SPRAVATO® (esketamine) nasal spray, CIII. Prescribing information. Janssen Pharmaceuticals, Inc.; 2025:16. Accessed May 13, 2025. https://www.janssenlabels.com/package-insert/product-monograph/prescribing-information/SPRAVATO-pi.pdf 3. Janik A et al. Poster presented at: Psych Congress; October 29-November 2, 2024; Boston, MA. Poster 112.
4. SPRAVATO® (esketamine) approved in the U.S. as the first and only monotherapy for adults with treatment-resistant depression. News release. Johnson & Johnson, Inc.; 2025. Accessed May 13, 2025. https://www.jnj.com/media-center/press-releases/spravato-esketamine-approved-in-the-u-s-as-the-first-and-only-monotherapy-for-adults-with-treatment-resistant-depression