Antipsychotic Utilization, Healthcare Resource Use, Costs, and Quality of Care in Patients With Schizophrenia Enrolled in Medicare, Medicaid, or Both Programs

Pengxiang Li[1], Zhi Geng[1,3], Carmela Benson[2], Jalpa A Doshi[1,3]

[1]University of Pennsylvania, Philadelphia, PA; [2]Johnson & Johnson, Titusville, NJ; [3]Leonard Davis Institute of Health Economics, Philadelphia, PA

This material is distributed for scientific purposes on J&J Medical Connect, and is not for promotional use

Background

- Schizophrenia is a serious chronic mental illness where adherence to antipsychotics (APs) is essential to reduce relapses and hospitalizations[1-3]
- Long-acting injectable antipsychotics (LAIs) are an important treatment option for improving adherence; real-world effectiveness studies show superiority of LAIs vs. traditional oral APs[4]
- Most U.S. patients with schizophrenia are publicly insured (Medicare, Medicaid, or dual eligible for both programs)[5]
 - For dual eligibles, Medicare is the primary payer including for prescription drugs
- Limited data exists on national rates and regional variation in AP use, healthcare resource use (HRU), costs, and quality of care across these three groups
 - Data is especially lacking in more recent years

Objective

- To provide national estimates and assess regional variations in AP utilization, treatment patterns, quality measures, HRU, and costs among patients with schizophrenia enrolled in Medicare only, Medicaid only, or both programs (dual eligible)

Methods

Data source

- 2023 Medicare Chronic Conditions Data Warehouse (CCW) 100% fee-for-service (FFS) files and the 2022 Transformed Medicaid Statistical Information System Analytic Files (TAF) files

Study sample

- Continuous fee-for-service Medicare Parts A, B, and D coverage in 2023 (Medicare only or dual eligible); continuous 12-month full Medicaid coverage in 2022 (Medicaid only)
- Age ≥18 years
- Evidence of ≥1 inpatient and/or ≥2 outpatient claims with a diagnosis for schizophrenia (ICD-10 codes F20.xx, F21 or F25.X) in the calendar year
- No evidence of ≥1 inpatient and/or ≥2 outpatient claims with a diagnosis for bipolar disorder (ICD-10 code F31.xx) in the calendar year
- Not missing state and county codes
- Residence in one of the 50 US states or D.C.

Outcome measures

Antipsychotic utilization

- All first-generation or second-generation AP: Any AP (LAI or OAP), any LAI, and OAP only
- Second-generation AP (SGA): Any AP, any LAI, and OAP only

Healthcare resource use

- All-cause, mental health–related, and schizophrenia-related inpatient visit, ER visit, and evaluation and management (E&M) visit

Healthcare costs

- All-cause, mental health–related and schizophrenia-related total costs, medical, and prescription costs

Quality measures

- AP adherence (defined as PDC ≥0.8)
- Inpatient readmission, outpatient E&M visit, any other outpatient service claim, AP dispensed within 30 days of discharge

Analysis

- Summary statistics on AP use, HRU, costs, and quality measures were reported at national, state, and county levels
- Regional variation was assessed using the coefficient of variation (CoV)

Results

- In a national sample of Medicare only (N=25,887), dual eligible (N=189,225), and Medicaid only (N=178,340) patients, the vast majority (80-88%) had evidence of AP use (**Figure 1**)
 - Less than one-quarter had evidence of LAI use (10% Medicare only, 20% dual eligible, and 27% Medicaid only)
 - Second-generation LAI use was even lower (6% Medicare only, 15% dual eligible, and 21% Medicaid only)
- Approximately one-quarter (22-26%) of patients had evidence of ≥1 hospitalization; rates of ER use were higher for Medicare only (40%) and dual eligibles (45%) compared to Medicaid only (25%) patients (**Table 1**)
- Mean total all-cause costs were $24,083 in Medicare only, $32,278 in dual eligibles, and $38,530 in Medicaid only patients during the calendar year (**Table 2**)
- Among patients with hospitalizations, around one-quarter (24-27%) had evidence of readmission within 30 days of discharge (**Figure 2**)
- Substantial geographic variation was observed within all three samples (use QR Code to see tables reporting state-level and county-level results)
 - Larger interstate variations were observed in any LAI use vs. any AP use (CoV: 0.39 vs. 0.04 for Medicare only, 0.22 vs. 0.03 for dual-eligible, and 0.25 vs. 0.12 for Medicaid only)
 - County-level variations were larger than state-level variations for all measures

Table 1: National Estimate of HRU

	Dual Eligible (Medicare + Medicaid)	Medicare Only	Medicaid Only
	% / Mean (SD)	% / Mean (SD)	% / Mean (SD)
N	189,225	25,887	178,340
All-cause HRU			
Any inpatient admission	26.1%	25.7%	22.3%
Number of inpatient admissions	1.9 (1.6)	1.8 (1.5)	1.8 (1.6)
Inpatient length of stay among those with inpatient admissions	24.4 (73.1)	20.6 (52.0)	23.8 (284.3)
Any ER visits	45.4%	40.3%	25.3%
Number of ER visits	3.9 (6.3)	3.5 (4.8)	3.9 (7.2)
Any E&M visit	82.8%	91.8%	40.0%
Mental health-related HRU			
Any inpatient admission	25.4%	24.6%	21.1%
Number of inpatient admissions	1.8 (1.5)	1.7 (1.3)	1.7 (1.5)
Inpatient length of stay among those with inpatient admissions	24.2 (72.1)	20.2 (52.6)	22.9 (292.4)
Any ER visits	35.0%	29.6%	14.5%
Number of ER visits	2.9 (4.7)	2.4 (3.1)	2.7 (4.1)
Any E&M visit	71.4%	84.0%	30.1%
Schizophrenia-related HRU			
Any inpatient admission	21.9%	19.2%	18.9%
Number of inpatient admissions	1.7 (1.3)	1.5 (1.0)	1.5 (1.2)
Inpatient length of stay among those with inpatient admissions	24.0 (75.9)	18.4 (56.3)	22.0 (302.0)
Any ER visits	25.8%	20.0%	7.9%
Number of ER visits	2.3 (3.5)	1.9 (2.1)	2.0 (2.5)
Any E&M visit	65.4%	75.9%	25.3%

Table 2: National Estimate of Costs

Costs	Dual Eligible (Medicare + Medicaid)	Medicare Only	Medicaid Only
	% / Mean (SD)	% / Mean (SD)	% / Mean (SD)
	189,225	25,887	178,340
All-cause costs			
All-cause Total costs	$32,278 ($43,579)	$24,083 ($37,913)	$38,530 ($48,001)
Medical costs	$17,444 ($35,400)	$16,215 ($32,691)	$32,020 ($45,574)
Inpatient costs	$8,078 ($25,111)	$7,701 ($23,391)	$4,461 ($19,107)
Emergency room (ER) costs	$698 ($2,129)	$539 ($1,666)	$335 ($1,670)
Outpatient costs	$8,668 ($18,516)	$7,974 ($16,358)	$27,225 ($40,709)
Pharmacy costs	$14,834 ($24,940)	$7,869 ($18,315)	$6,511 ($14,190)
Mental health-related costs			
Mental-health related Total costs	$19,665 ($30,315)	$13,812 ($26,071)	$21,406 ($40,340)
Medical costs	$11,385 ($25,303)	$9,880 ($2,2912)	$16,938 ($38,759)
Inpatient costs	$7,594.45 ($22,784)	$6925 ($20,978)	$3806 ($16,664)
Emergency room (ER) costs	$370 ($1,362)	$252 ($1,100)	$138 ($914)
Outpatient costs	$3,420 ($6,780)	$2703 ($5179)	$12,995 ($34,832)
Pharmacy costs	$8,281 ($17,668)	$3,932 ($13,090)	$4,468 ($10,599)
Schizophrenia-related costs			
Schizophrenia-related Total costs	$14,654 ($24,561)	$8,995 ($19,038)	$14,395 ($26,646)
Medical costs	$8,579 ($2,1420)	$6,452 ($17,334)	$10,461 ($24,819)
Inpatient costs	$6,040 ($19,619)	$4,569 ($16,062)	$3,100 ($14,725)
Emergency room (ER) costs	$201 ($907)	$125 ($809)	$59 ($533)
Outpatient costs	$2,338 ($5,731)	$1,758 ($4,238)	$7,302 ($19,863)
Pharmacy costs	$6,075 ($12,536)	$2,542 ($8,063)	$3934 ($9,059)

Figure 1: National Estimate of Antipyschotic Use

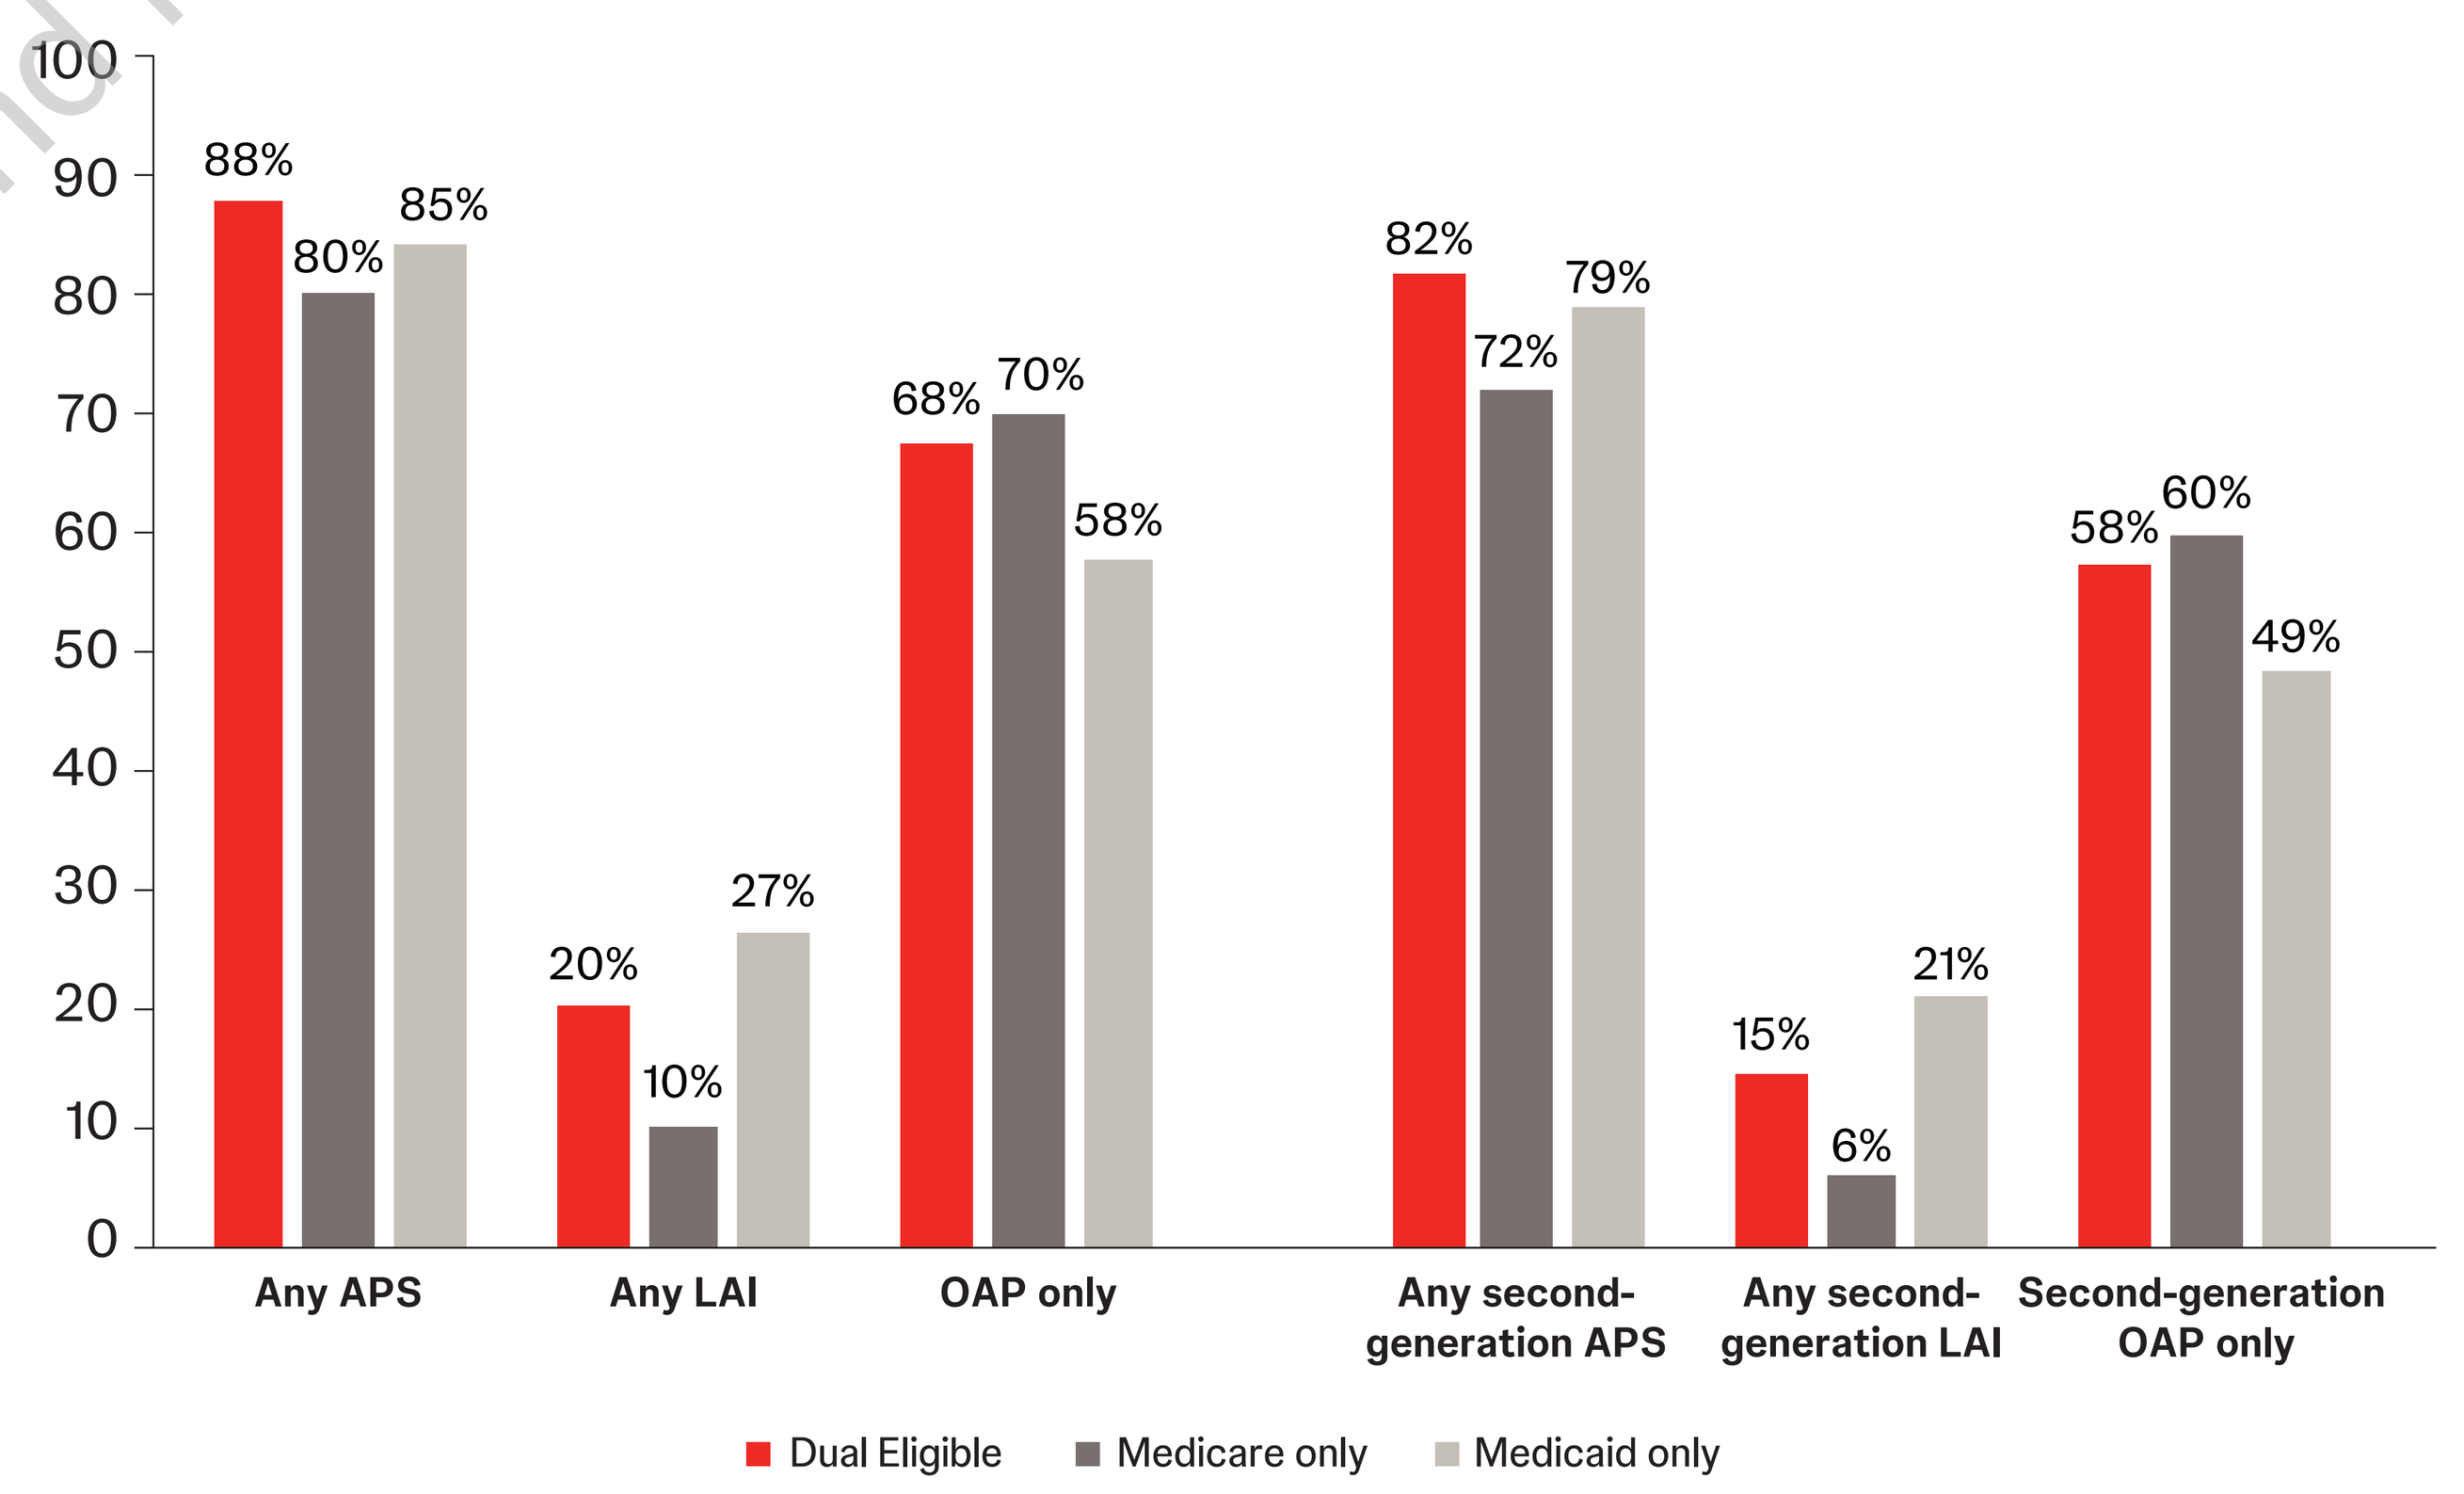
100
90
80
70
60
50
40
30
20
10
0
88%
80%
85%
20%
10%
27%
68%
70%
58%
82%
72%
79%
15%
6%
21%
58%
60%
49%
Any APS
Any LAI
OAP only
Any second-generation APS
Any second-generation LAI
Second-generation OAP only
Dual Eligible
Medicare only
Medicaid only

Figure 2: National Estimate of Quality Measures

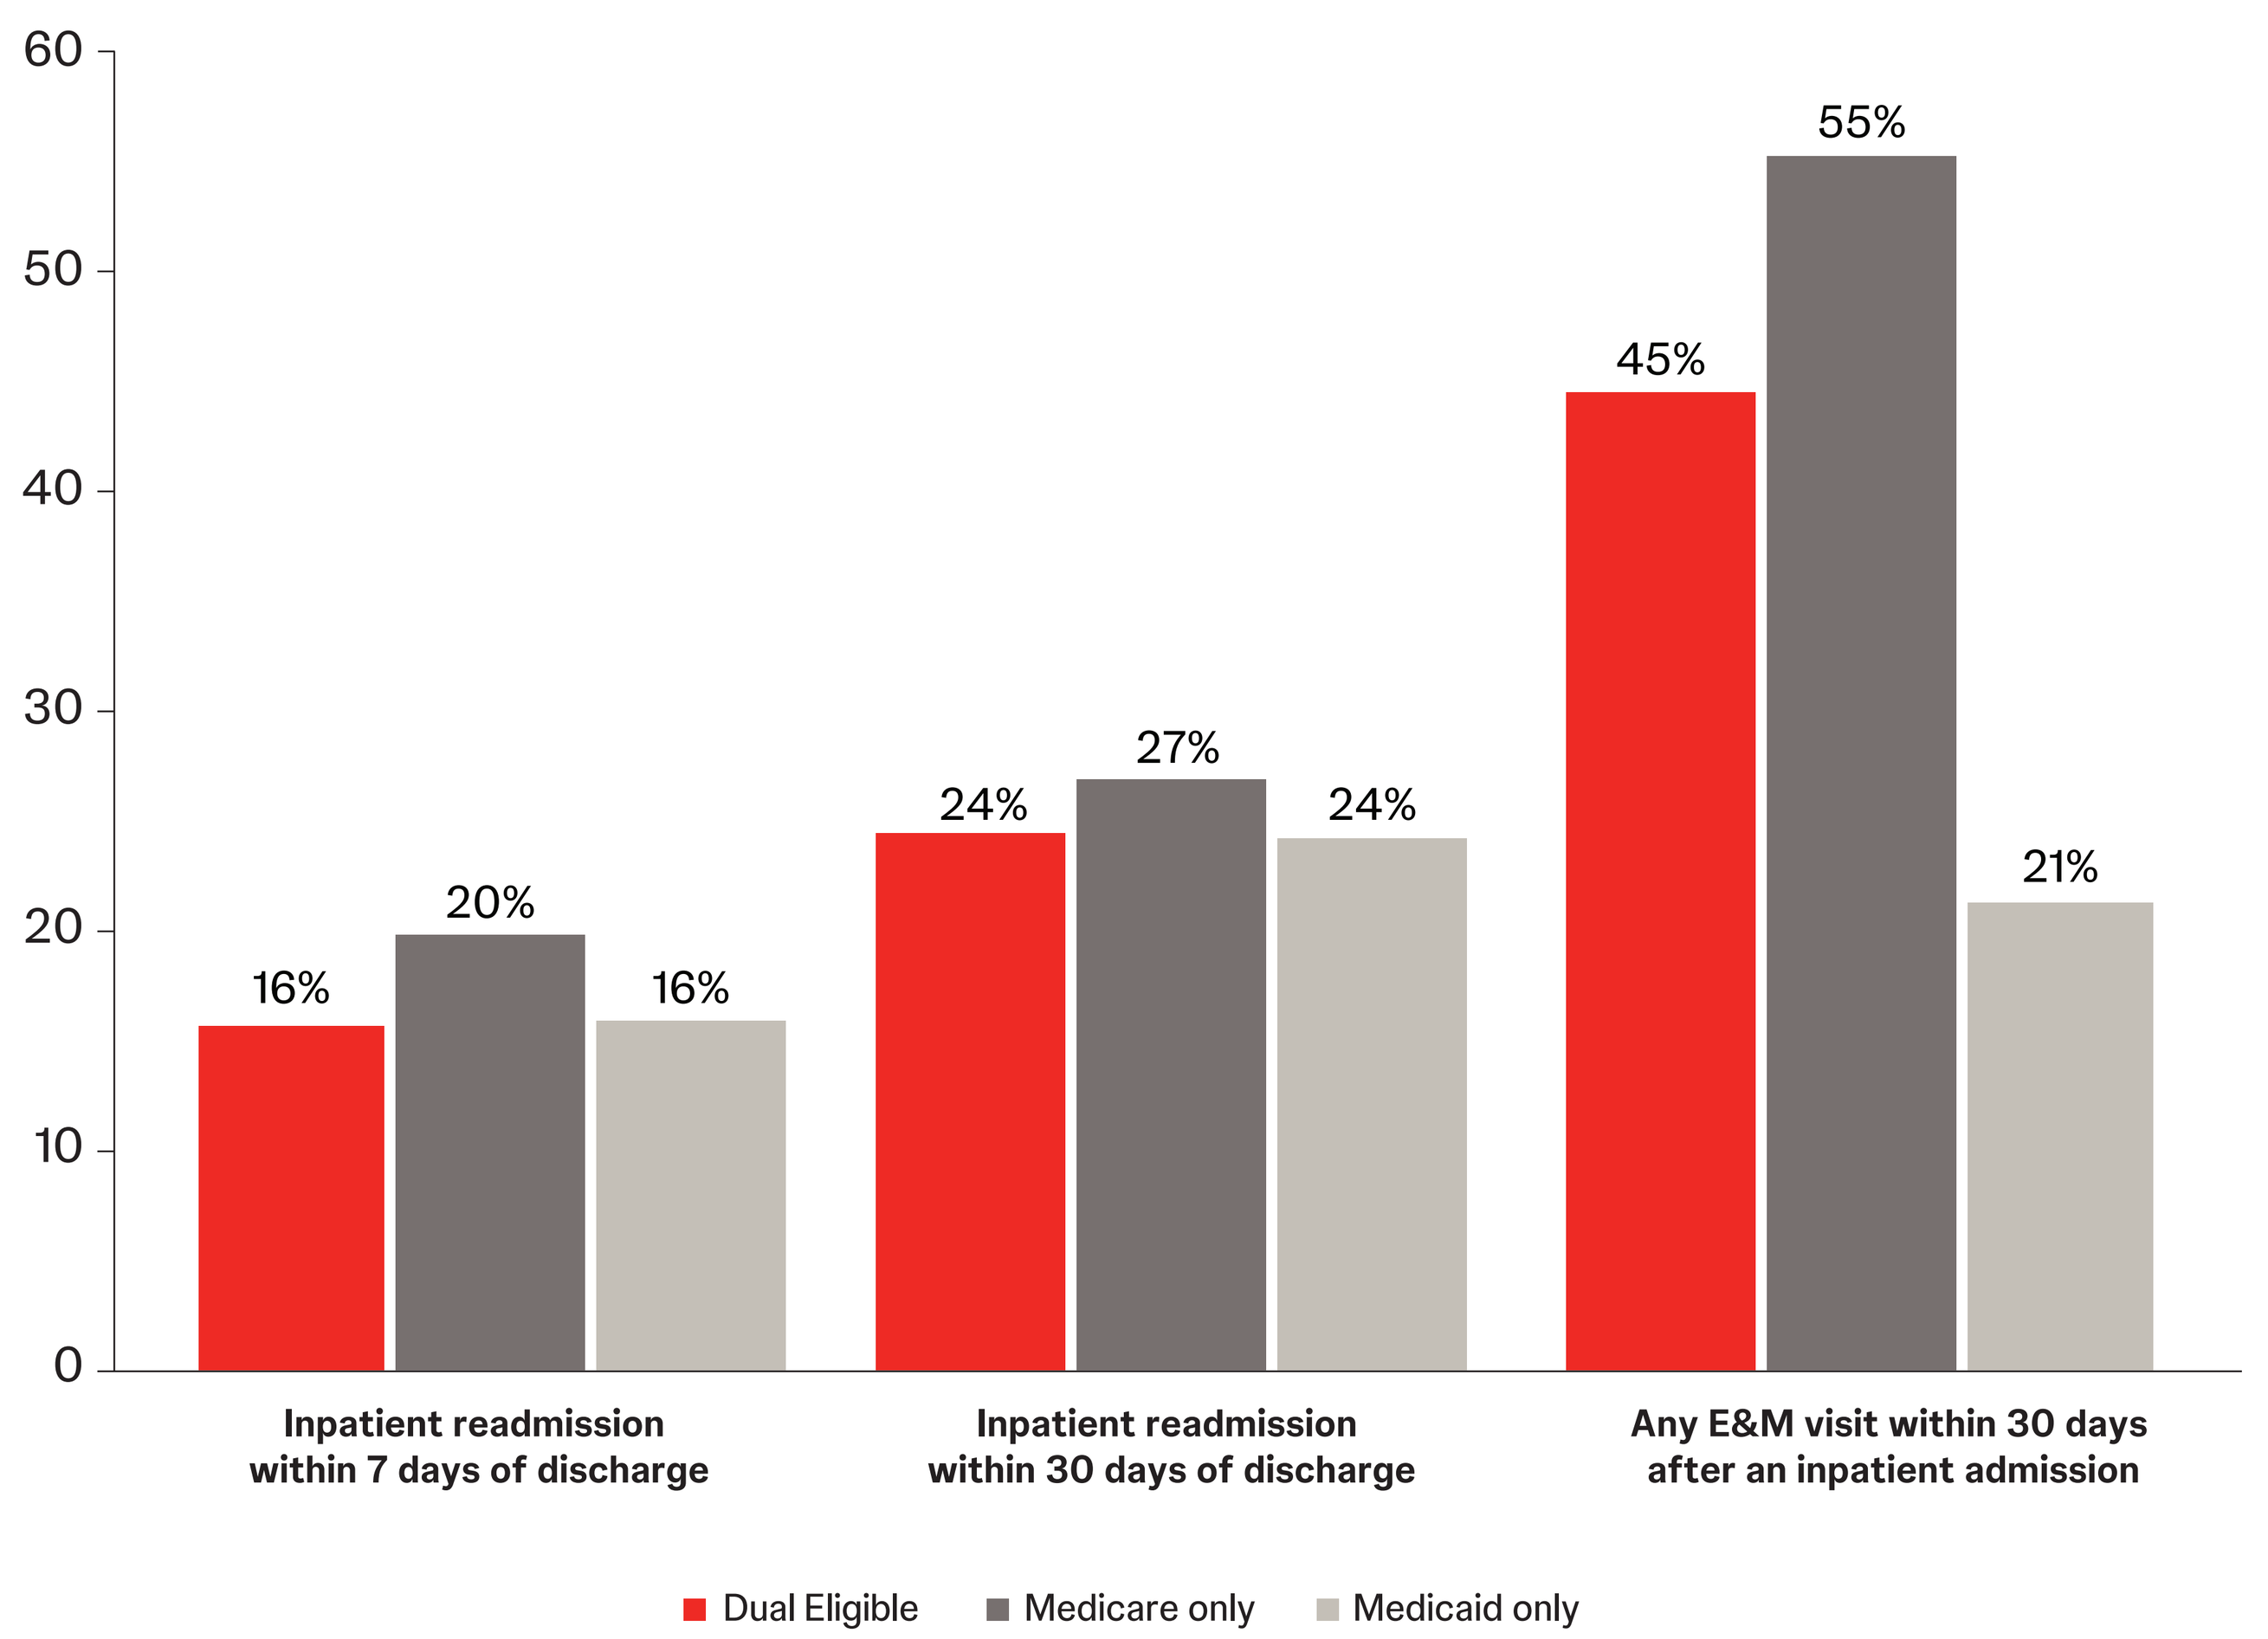
60
50
40
30
20
10
0
16%
20%
16%
24%
27%
24%
45%
55%
21%
Inpatient readmission within 7 days of discharge
Inpatient readmission within 30 days of discharge
Any E&M visit within 30 days after an inpatient admission
Dual Eligible
Medicare only
Medicaid only

The rate of Antipsychotic dispensed within 30 days after an inpatient admission was 58.8% for Duals and Medicaid only and 47.2% for Medicare only.

Conclusions

- This national study of Medicare only, Medicaid only, and dually eligible patients found low utilization of LAIs and high levels of hospital admissions/readmissions, ER visits, and costs
- Substantial state- and county-level variation in these measures was observed
- Our findings suggest an inefficiency and ongoing unmet need despite currently available AP treatments

Disclosures

CB is an employee and shareholder of Johnson & Johnson.

Neuropsychiatry

Scan the QR code

The QR code is intended to provide scientific information for individual reference, and the information should not be altered or reproduced in any way.

This study was funded by Johnson & Johnson.

Presented at Neuroscience Education Institute (NEI) Fall Congress; November 6-9, 2025; Colorado Springs, Colorado, USA

References

1. American Psychiatric Association, DSM-5 (2013). 2. Olivares JM, et al. *Ann Gen Psychiatry*. 2013 Oct 23;12(1):32. 3. Perkins AJ, et al. *BMC Psychiatry*. 2023;23(1):64. 4. Li et al. *Adv Ther*. 2025;42(2):1251-1264. 5. Li P, et al. *J Med Econ*. 2023 Jan-Dec;26(1):525-536.