280

An Economic Model Assessing Costs, Relapses, and Antipsychotic Adherence of Switching Medicaid Beneficiaries With Schizophrenia From Oral Antipsychotics to Once-Monthly, Once-Every-Three-Months, and Once-Every-Six-Months Paliperidone Palmitate

Carmela Benson, MSc[1], Laura Morrison, MScPH[2], Béatrice Libchaber, MA[2], Lilian Diaz, MScPH[2], Yuxian Du, PhD[1], Dominic Pilon, MA[2]

[1]Johnson & Johnson, Titusville, NJ, USA, [2]Analysis Group ULC, Montreal, Quebec, Canada

Background

- Schizophrenia is associated with substantial clinical and economic burden in the United States (US), with an estimated prevalence ranging from 1.46% to 4.23% among Medicaid beneficiaries across 7 states and total excess costs for patients with schizophrenia, relative to those without, of more than $340 billion in the US in 2019[1-3]
- The large clinical and economic burden of schizophrenia highlights the importance of long-term compliance to antipsychotic (AP) treatment to improve symptoms and reduce the likelihood of relapse[4]
- Long-acting injectable (LAI) APs, including paliperidone palmitate, have been shown to be associated with improved AP adherence and clinical outcomes compared to oral APs (OAPs), while remaining cost-neutral[5-8]
- In an evolving LAI treatment space with emergent real-world data, there is a need to better understand the impact of paliperidone palmitate treatment pathways on economic and clinical outcomes among a population of Medicaid beneficiaries with schizophrenia

Objective

- To evaluate the plan-level impact on healthcare costs, relapse rates, readmission rates, and average AP adherence of switching Medicaid beneficiaries with schizophrenia from OAPs to once-monthly paliperidone palmitate (PP1M) alone, and with subsequent transitions to once-every-three-months (PP3M) and once-every-six-months paliperidone palmitate (PP6M)

Methods

Model framework/structure

- A Markov model with twelve 3-month cycles over a 36-month time frame was developed from a Medicaid payer perspective (**Figure 1**)
- Three cohorts of adults with schizophrenia were included: those treated with once- or twice-daily OAPs, those switching to PP1M only, and those switching to PP1M with subsequent on-label transitions to PP3M and PP6M
- For the switching cohorts, 20% of the population was switched to PP1M, based on real-world evidence of LAI utilization rates among Medicaid beneficiaries[9]
- The model evaluated and compared total plan-level healthcare costs (based on costs of AP treatment and relapse, reported in 2025 US dollars [USD]), relapse rates, readmission rates, and average AP adherence among the three cohorts of patients

Model assumptions

- At each 3-month cycle, patients may or may not be adherent to AP treatment, may or may not experience a relapse, and could continue receiving the same AP treatment or switch treatment
- The risk of relapse differed according to treatment and adherence status, and medication switch rates differed according to relapse and adherence status; patients could have experienced multiple relapses and treatment switches (up to 12) over the 36-month model time horizon
- Patients transitioning from PP1M to PP3M/PP6M had to do so on-label (i.e., after two 3-month cycles of PP1M, at the correct corresponding dose strength, with no evidence of relapse during the current cycle)[10,11]
- Patients on PP3M/PP6M could switch to another paliperidone palmitate formulation, or an OAP, while patients on an OAP could only switch to another OAP
- Patients switching to PP1M were assumed to be part of the PP1M treatment arm regardless of whether they switched to an OAP over the course of the model

Model inputs

- All model inputs including population inputs, rates of relapse, readmission, adherence, and treatment switches, as well as costs of treatment and relapse, were literature-based[1,9,12-18]
- Considering both statutory and inflation discounts, a 70% discount was used for PP1M and PP3M treatment costs; for PP6M, given that it is a newer product, a 60% discount was applied, in line with prior literature[12,19]

Figure 1: Model structure

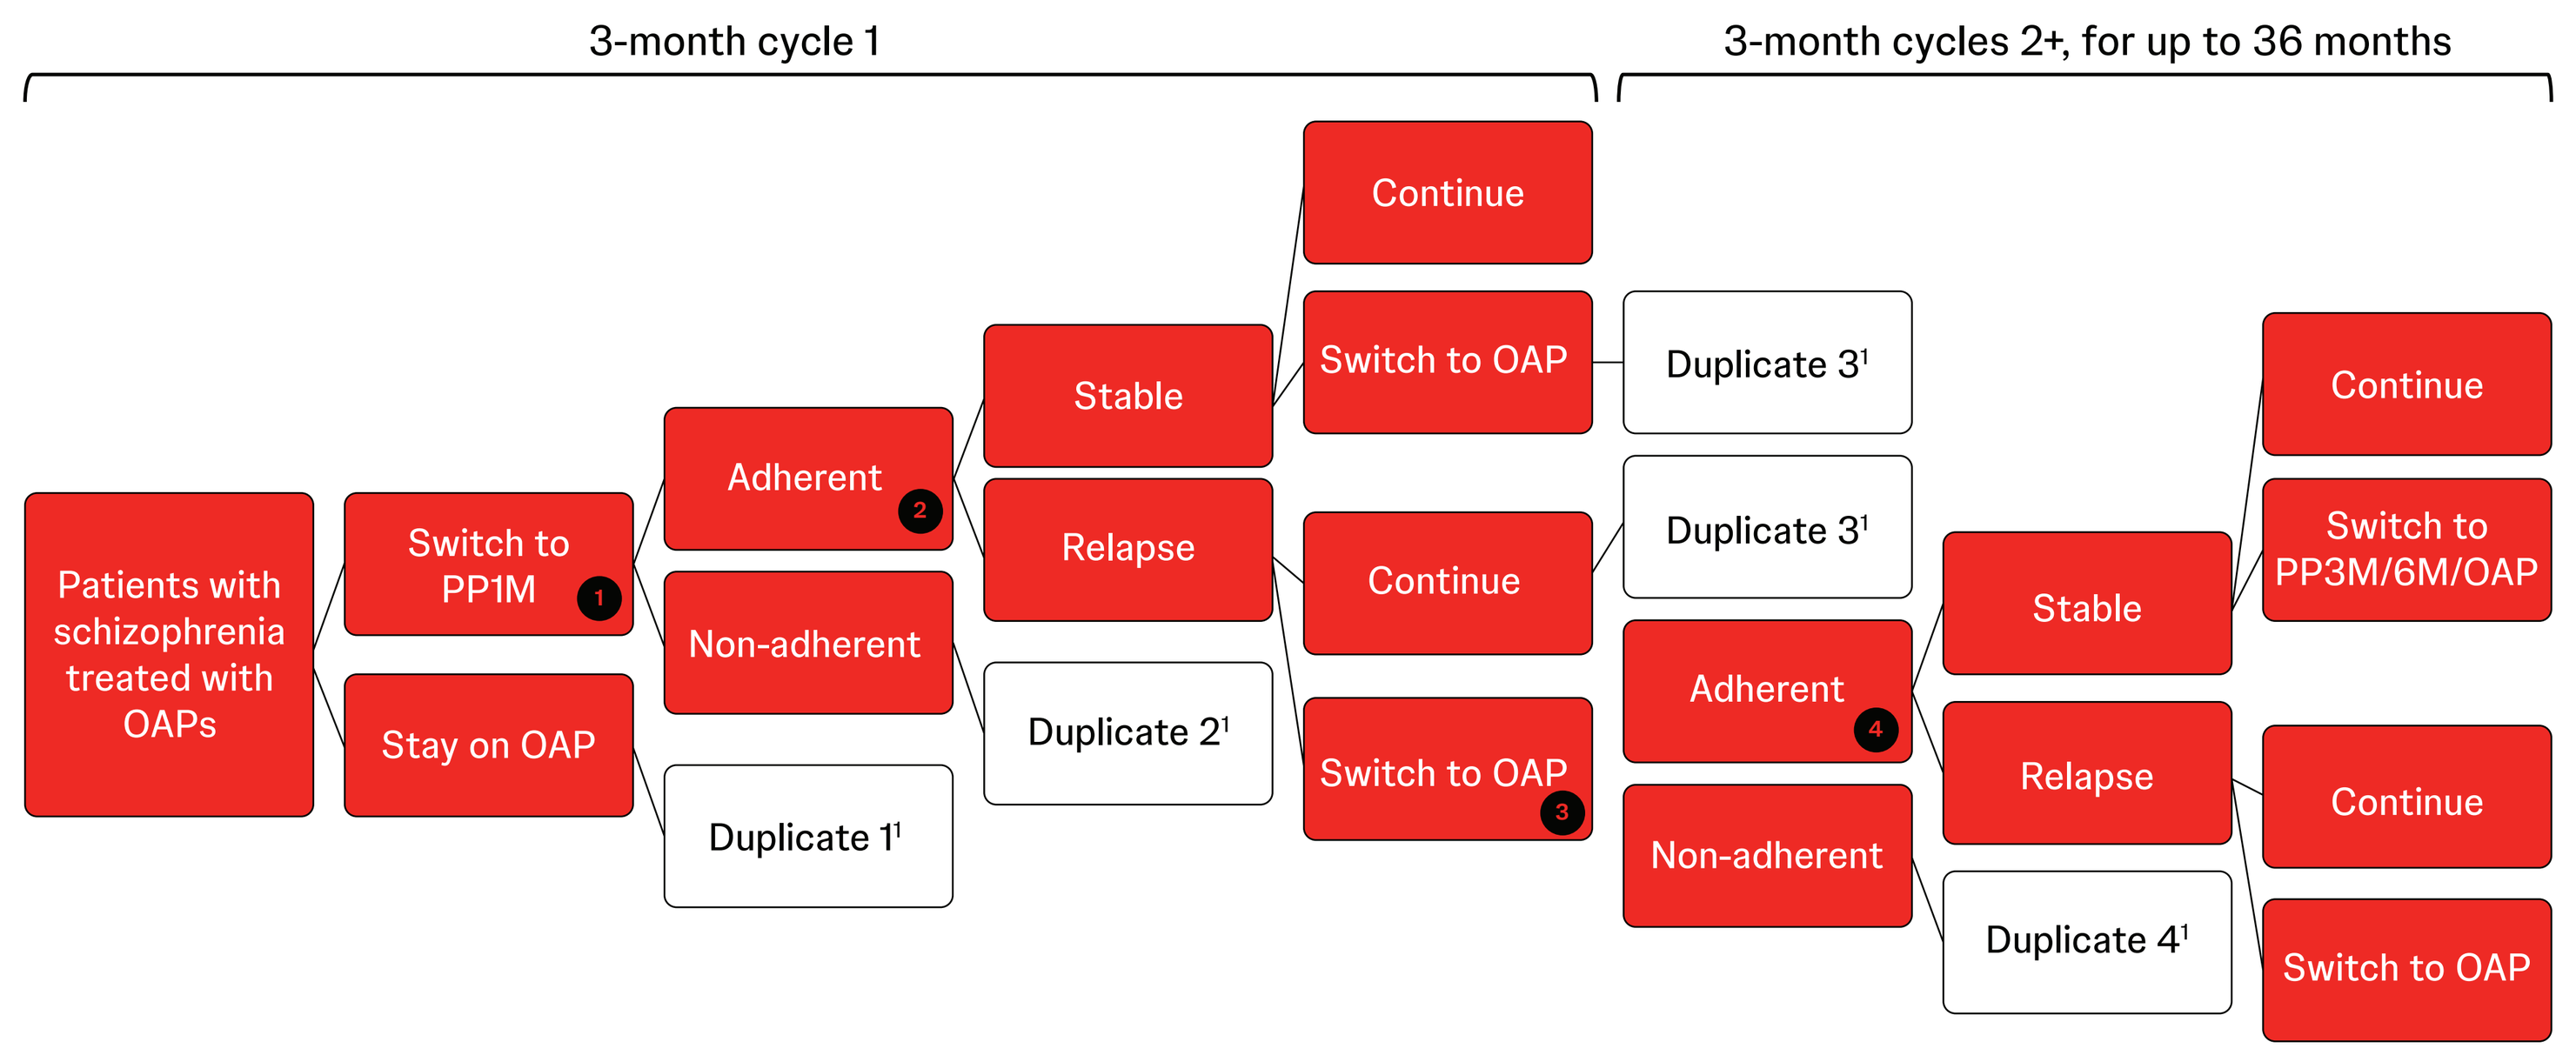
3-month cycle 1
3-month cycles 2+, for up to 36 months
Patients with schizophrenia treated with OAPs
Switch to PP1M
1
Stay on OAP
Adherent
2
Non-adherent
Duplicate 1¹
Stable
Relapse
Duplicate 2¹
Continue
Switch to OAP
Continue
Switch to OAP
3
Duplicate 3¹
Duplicate 3¹
Adherent
4
Non-adherent
Stable
Relapse
Duplicate 4¹
Continue
Switch to PP3M/6M/OAP
Continue
Switch to OAP

OAP = oral antipsychotic; PP1M = once-monthly paliperidone palmitate; PP3M = once-every-three-months paliperidone palmitate; PP6M = once-every-six-months paliperidone palmitate.
[1]At this point the branch is being duplicated from the corresponding number. Duplication does not include any switches to PP3M and PP6M from the OAP cohort.

Results

- In a hypothetical health plan of 1 million Medicaid beneficiaries, an estimated 13,419 adult members with schizophrenia treated with OAPs incurred plan-level costs of $1,253.5 million, based on 33,671 relapses, 7,778 readmissions, and average AP adherence of 69.5% over the 36-month model time horizon (**Table 1**)

Table 1: Plan-level costs, relapses, readmissions, and average AP adherence associated with switching 20% of Medicaid beneficiaries with schizophrenia from OAPs to PP1M, with subsequent transitions to PP3M and PP6M

	Patients on any OAPs [A]	Patients switching to PP1M only [B]	Difference [B – A]	Patients switching to PP1M, PP3M, and PP6M [C]	Difference [C – A]
Plan-level costs (2025 USD)					
Year 1	$417,824,890	$379,828,955	–$37,995,934	$378,754,322	–$39,070,567
Net cost per patient switched per year	-	-	–$14,158	-	–$14,558
Year 2	$417,824,890	$381,940,519	–$35,884,370	$377,788,663	–$40,036,226
Net cost per patient switched per year	-	-	–$13,371	-	–$14,918
Year 3	$417,824,890	$385,536,042	–$32,288,848	$380,857,921	–$36,966,969
Net cost per patient switched per year	-	-	–$12,031	-	–$13,774
Total	**$1,253,474,669**	**$1,147,305,517**	**–$106,169,152**	**$1,137,400,906**	**–$116,073,763**
Net cost per patient switched over 3 years	-	-	–$39,559	-	–$43,250
Relapses					
Year 1	11,224	9,614	–1,610	9,545	–1,678
Year 2	11,224	9,782	–1,442	9,517	–1,706
Year 3	11,224	9,926	–1,297	9,609	–1,614
Total	**33,671**	**29,322**	**–4,349**	**28,672**	**–4,999**
Quality measures					
Readmissions					
Year 1	2,593	2,221	–372	2,205	–388
Year 2	2,593	2,260	–333	2,198	–394
Year 3	2,593	2,293	–300	2,220	–373
Total	**7,778**	**6,773**	**–1,005**	**6,623**	**–1,155**
Average AP adherence[1]					
Year 1	69.5%	78.9%	9.4%	82.5%	13.0%
Year 2	69.5%	76.3%	6.8%	86.8%	17.3%
Year 3	69.5%	75.6%	6.1%	86.8%	17.3%
Average	**69.5%**	**76.9%**	**7.4%**	**85.4%**	**15.9%**

AP = antipsychotic; OAP = oral antipsychotic; PP1M = once-monthly paliperidone palmitate; PP3M = once-every-three-months paliperidone palmitate; PP6M = once-every-six-months paliperidone palmitate; USD = United States Dollars.
[1]Average AP adherence was reported among patients who switched and was compared between the cohort of patients remaining on OAPs relative to the 20% of patients who switched to PP1M, as well as the 20% of patients with subsequent on-label transitions to PP3M and PP6M.

Plan-level costs

- Switching 20% (n=2,684) of patients from OAPs to PP1M only resulted in plan-level cost savings of $106.2 million (**Figure 2**)
- Incorporating subsequent transitions to PP3M and PP6M led to additional plan-level savings of $9.9 million relative to patients treated with PP1M only, representing $116.1 million in plan-level cost savings relative to patients treated with OAPs

Relapses and Readmissions

- Switching 20% of patients from OAPs to PP1M only avoided 4,349 relapses, with subsequent transitions to PP3M and PP6M avoiding an additional 650 relapses (**Figure 3**)
- Similarly, switching patients from OAPs to PP1M avoided 1,005 readmissions, and subsequent transitions to PP3M and PP6M additionally avoided 150 readmissions

Adherence

- Among the 20% of patients who switched from OAPs to PP1M only, switching to PP1M resulted in a 7.4 percentage point increase in average adherence to any AP, while incorporating transitions to PP3M and PP6M resulted in an additional 8.5 percentage point increase in average AP adherence (**Figure 4**)

Figure 2: Total plan-level costs and cost savings associated with switching 20% of Medicaid beneficiaries with schizophrenia from OAPs to PP1M, with subsequent transitions to PP3M and PP6M

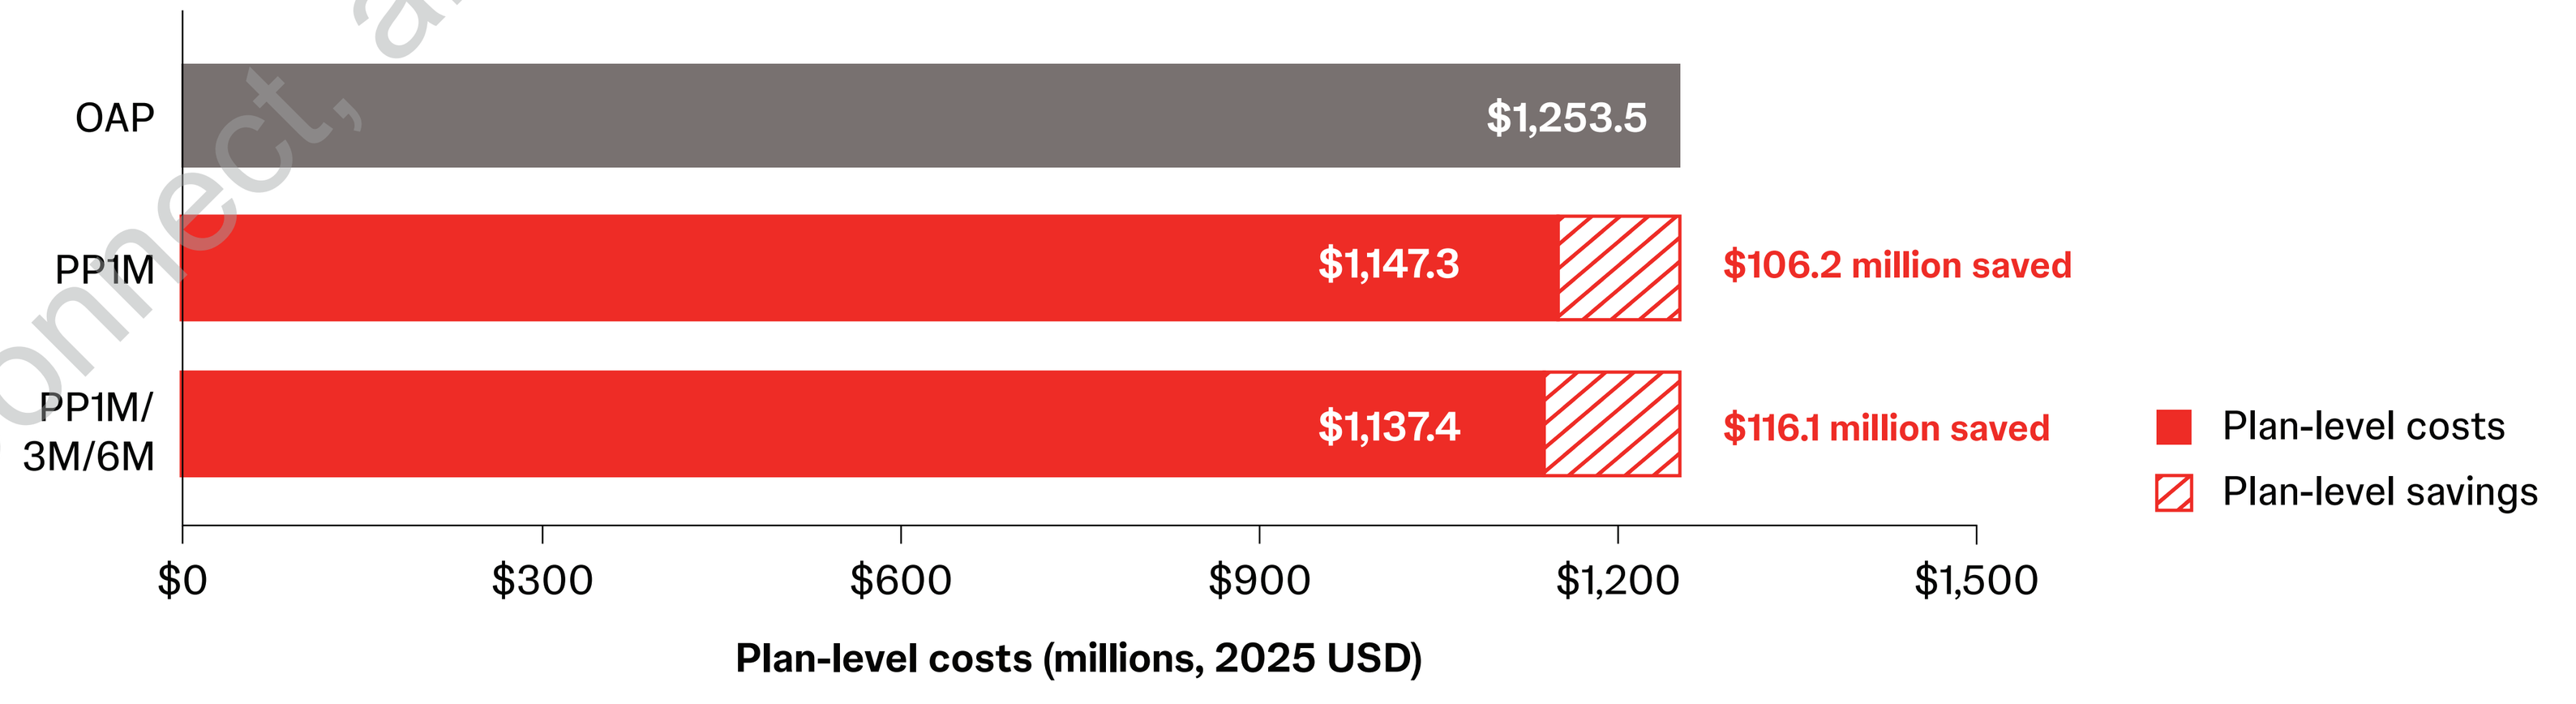
OAP
PP1M
PP1M/ 3M/6M
$1,253.5
$1,147.3
$1,137.4
$106.2 million saved
$116.1 million saved
Plan-level costs
Plan-level savings
$0
$300
$600
$900
$1,200
$1,500
Plan-level costs (millions, 2025 USD)

OAP = oral antipsychotic; PP1M = once-monthly paliperidone palmitate; PP3M = once-every-three-months paliperidone palmitate; PP6M = once-every-six-months paliperidone palmitate; USD = United States Dollars.

Figure 3: Total relapses and relapses avoided associated with switching 20% of Medicaid beneficiaries with schizophrenia from OAPs to PP1M, with subsequent transitions to PP3M and PP6M

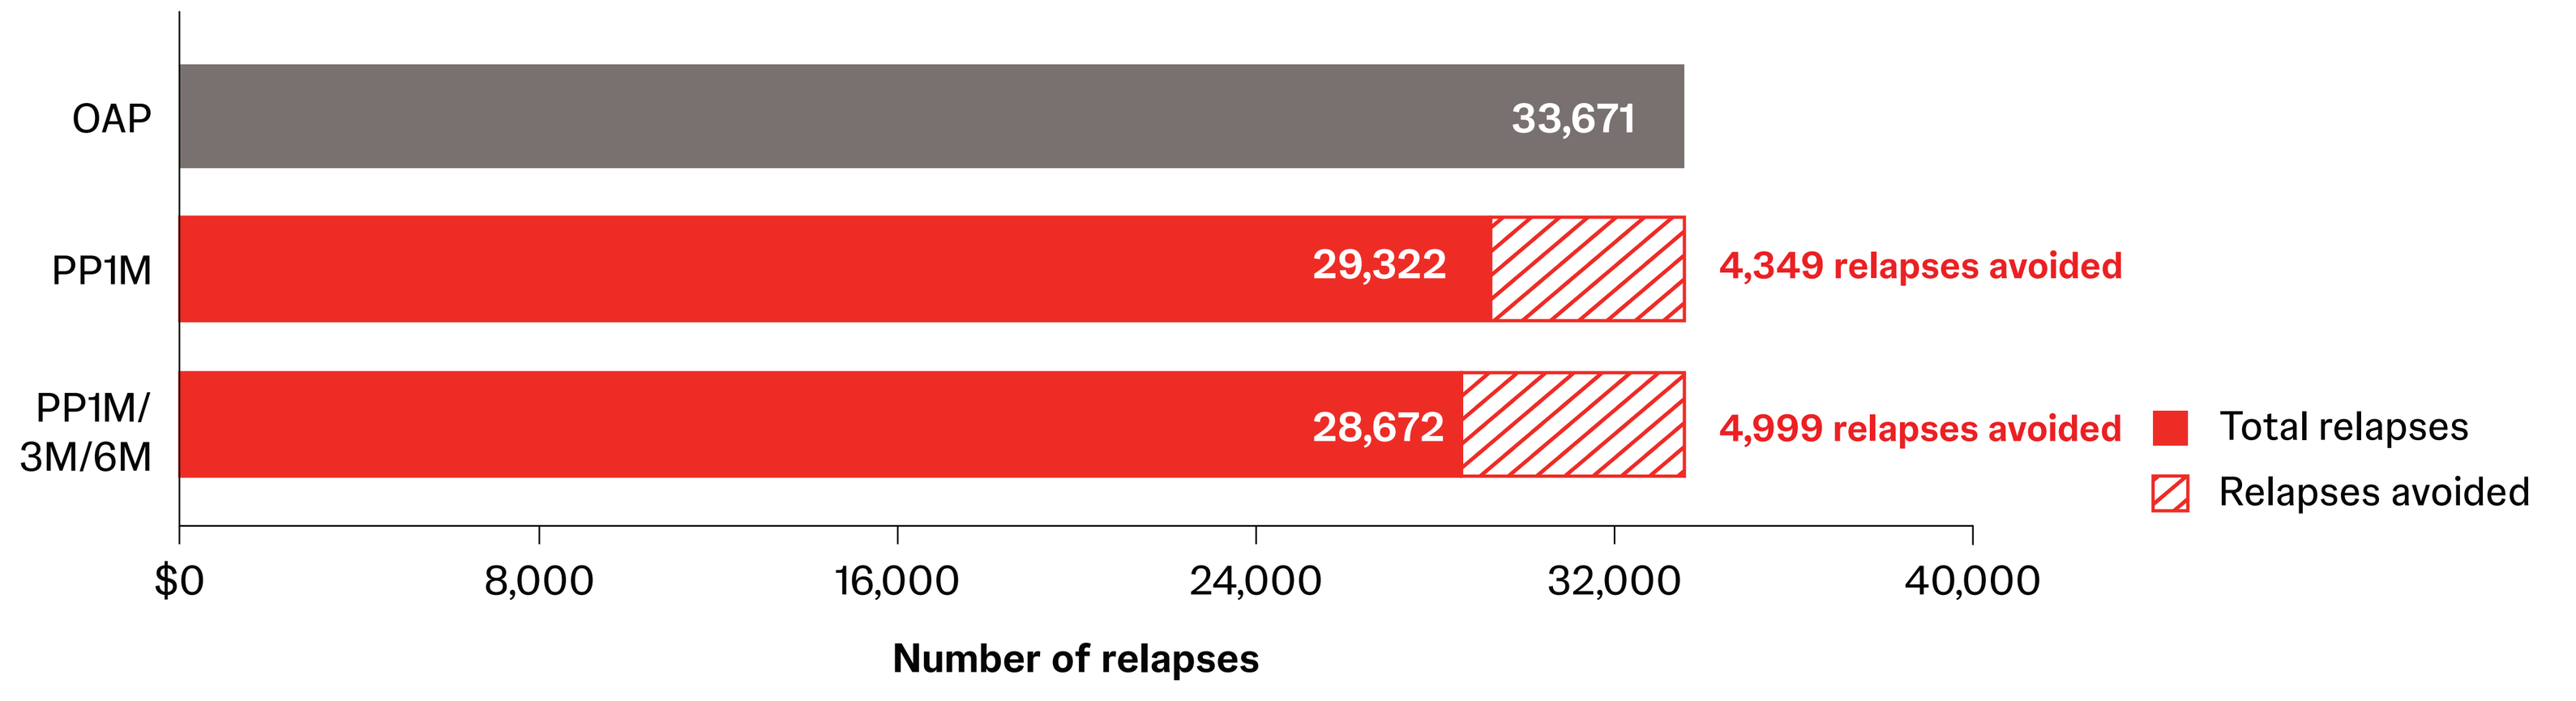
OAP
PP1M
PP1M/ 3M/6M
33,671
29,322
28,672
4,349 relapses avoided
4,999 relapses avoided
Total relapses
Relapses avoided
$0
8,000
16,000
24,000
32,000
40,000
Number of relapses

OAP = oral antipsychotic; PP1M = once-monthly paliperidone palmitate; PP3M = once-every-three-months paliperidone palmitate; PP6M = once-every-six-months paliperidone palmitate.

Figure 4: Average AP adherence associated with switching 20% of Medicaid beneficiaries with schizophrenia from OAPs to PP1M, with subsequent transitions to PP3M and PP6M

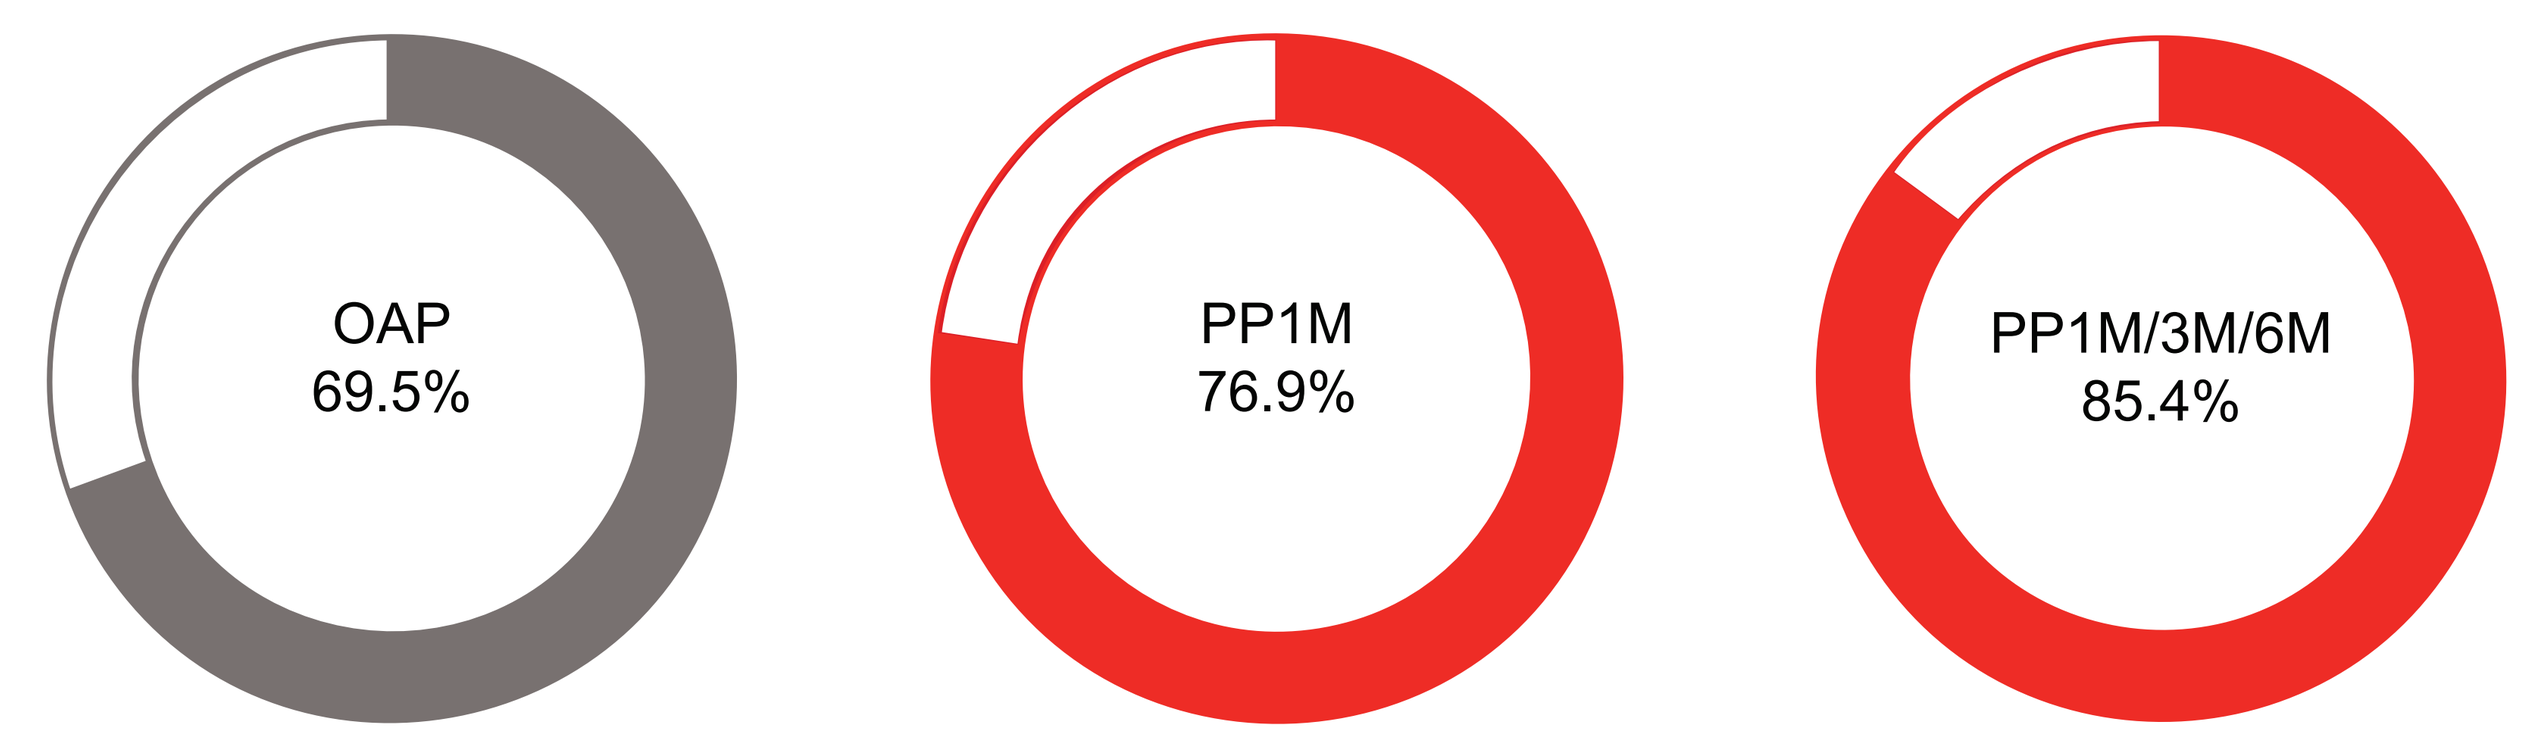
OAP
69.5%
PP1M
76.9%
PP1M/3M/6M
85.4%

AP = antipsychotic; OAP = oral antipsychotic; PP1M = once-monthly paliperidone palmitate; PP3M = once-every-three-months paliperidone palmitate; PP6M = once-every-six-months paliperidone palmitate.

Limitations

- Results reported are specific to patients with schizophrenia covered by Medicaid and may not be generalizable to patients with other types of insurance or those who are uninsured
- Furthermore, as the estimated number of adult Medicaid beneficiaries with schizophrenia in a 1 million member plan is based on single-state prevalence data, plan-level cost savings and relapses avoided may vary if national prevalence estimates differ
- Model inputs were based on data available in published literature and may not fully represent outcomes observed in clinical practice; analyses based on real-world data are warranted to confirm the findings of this study
- Given that PP6M is a newer product, the treatment discount was based on assumptions, and real-world rebates may vary widely

Conclusions

- Based on the findings of this 36-month Markov model, switching patients from OAPs to PP1M is associated with notable plan-level cost savings driven by avoided relapses and increased adherence to APs
- These results highlight the economic benefit of paliperidone palmitate LAIs, with longer-acting formulations PP3M and PP6M leading to additional incremental cost savings, further reductions in relapse rates, and continued AP adherence improvements
- This study offers important insights that should be taken into account in real-world clinical practice to inform treatment decisions

Disclosures

CB and YD are employees of Johnson & Johnson. LM, BL, LD, and DP are employees of Analysis Group, ULC., a consulting company that has provided paid consulting services to Johnson & Johnson which funded the development and conduct of this study.

Neuropsychiatry

Scan the QR code

The QR code is intended to provide scientific information for individual reference, and the information should not be altered or reproduced in any way.

This study was funded by Johnson & Johnson

Presented at AMCP Nexus 2025; October 27-30, 2025; National Harbor, Maryland, USA

References
1. Finnerty MT, et al. *Schizophrenia (Heidelb)*. 2024;10(1):68. 2. Kadakia A, et al. *J Clin Psychiatry*. 2022;83(6):22m14458. 3. Pilon D, et al. *Curr Med Res Opin*. 2021; 37(10):1811-1819. 4. Keepers GA, et al. *Am J Psychiatry*. 2020;177(9):868-872. 5. Lin D, et al. *CNS Drugs*. 2021;35(5):469-481. 6. Pilon D, et al. *Clin Ther*. 2017;39(10):1972-1985 e1972. 7. El Khoury AC, et al. *Curr Med Res Opin*. 2019;35(3):395-405. 8. Patel C, et al. *Drugs Real World Outcomes*. 2020;7(1):19-29. 9. Patel C, et al. *J Med Econ*. 2022;25(1):792-807. 10. INVEGA HAFYERA® (paliperidone palmitate) extended-release injectable suspension, for intramuscular use [highlights of prescribing information]. Titusville, NJ: Janssen Pharmaceuticals, Inc; October 2021. 11. INVEGA TRINZA® (paliperidone palmitate) extended-release injectable suspension, for intramuscular use [highlights of prescribing information]. Titusville, NJ: Janssen Pharmaceuticals, Inc. January 2019. 12. Morrison L, et al. *J Manag Care Spec Pharm*. 2023;29(2):161-171. 13. Centers for Medicare & Medicaid Services. ASP Pricing Files. March 2025. 14. Healthcare Cost and Utilization Project (HCUP). Statistical brief #307: Clinical Conditions With Frequent, Costly Hospital Readmissions by Payer, 2020. 2020. 15. Healthcare Cost and Utilization Project (HCUP). Readmission Stays by Type, United States, 2021 Graphed: Average hospital costs per stay. 2021. 16. Zacker C, et al. *Clinicoecon Outcomes Res*. 2024;16:567-579. 17. Patel C, et al. Real-World Adherence, Healthcare Resource Utilization, and Costs Among Patients With Schizophrenia Utilizing Once-Monthly, Once-Every-Three-Months, and Once-Every-Six-Months Paliperidone Palmitate in the United States. International Society for Pharmacoeconomics and Outcomes Research (ISPOR) 2025. 18. Healthcare Cost and Utilization Project (HCUP). Weighted national estimates from HCUP National (Nationwide) Inpatient Sample (NIS). 19. Kelly C. Price Inflation Rebates and the Spectre of 100% Discounts in Medicare Part D. January 21, 2020.

This material is distributed for scientific purposes on J&J Medical Connect, and is not for promotional use