Efficacy and Safety of Esketamine Nasal Spray as Monotherapy in Adults with Treatment-Resistant Depression: A Randomized, Double-Blind, Placebo-Controlled Study

Adam Janik,[1] Xin Qiu,[2] Rosanne Lane,[3] Vanina Popova,[4] Wayne C. Drevets,[1] Carla M. Canuso,[3] Dong Jing Fu[3]

[1]Janssen Research & Development, LLC, San Diego, CA, USA; [2]Janssen Research & Development, LLC, Raritan, NJ, USA; [3]Janssen Research & Development, LLC, Titusville, NJ, USA; [4]Janssen Pharmaceutica NV, Beerse, Belgium

Background

- Treatment-resistant depression (TRD) is a chronic condition and is associated with higher rates of relapse, increased mortality, and a greater risk for suicide compared to non-treatment-resistant depression.[1]
- Esketamine (ESK) nasal spray is approved in 75 countries for use in conjunction with an oral antidepressant for TRD.[2,3]
- Given that ESK is only approved for use in conjunction with an oral antidepressant, obtaining approval to use ESK as monotherapy would be an important development for clinical practice.
- This was the first Phase 4 study to demonstrate efficacy and safety of ESK nasal spray as a monotherapy for TRD.

Objective

- To assess efficacy and safety of 2 fixed doses (56 mg and 84 mg) of ESK nasal spray monotherapy compared with placebo (PBO) in reducing depressive symptoms in adults with TRD.

Methods

Study participants

Inclusion criteria

- Adult participants ≥18 years of age
- Recurrent or single (duration ≥2 years) episode of MDD (per DSM-5 criteria), without psychotic features*
- Medically stable
- Non-response# (≤25% improvement) to ≥2 oral antidepressants used during the current depressive episode
- IDS-C_{30} total score of ≥34

Exclusion criteria

- The participant has used ketamine/ESK (lifetime)
- Previous non-responsiveness to ECT in the current MDD episode (at least 7 treatments with unilateral/bilateral ECT)
- Participant underwent vagal nerve or deep brain stimulation in current depression episode
- Participant with anatomical or medical condition that may impede delivery or absorption of nasal spray study drug
- Homicidal/suicidal ideation/intent within 6 months, or suicidal behavior within the past year pre-screening
- Moderate/severe substance or alcohol use disorder (DSM-5 criteria), except nicotine or caffeine, within 6 months pre-screening

*Based upon clinical assessment and confirmed by the Mini International Neuropsychiatric Interview. #Non-response to oral antidepressants was assessed using MGH-ATRQ. DSM-5, Diagnostic and Statistical Manual of Mental Disorders (5th edition); ECT, electroconvulsive therapy; ESK, esketamine; IDS-C30, Inventory of Depressive Symptomatology-Clinician rated, 30-item; MDD, major depressive disorder; MGH-ATRQ, Massachusetts General Hospital-Antidepressant Treatment Response Questionnaire.

FIGURE 1. Study design

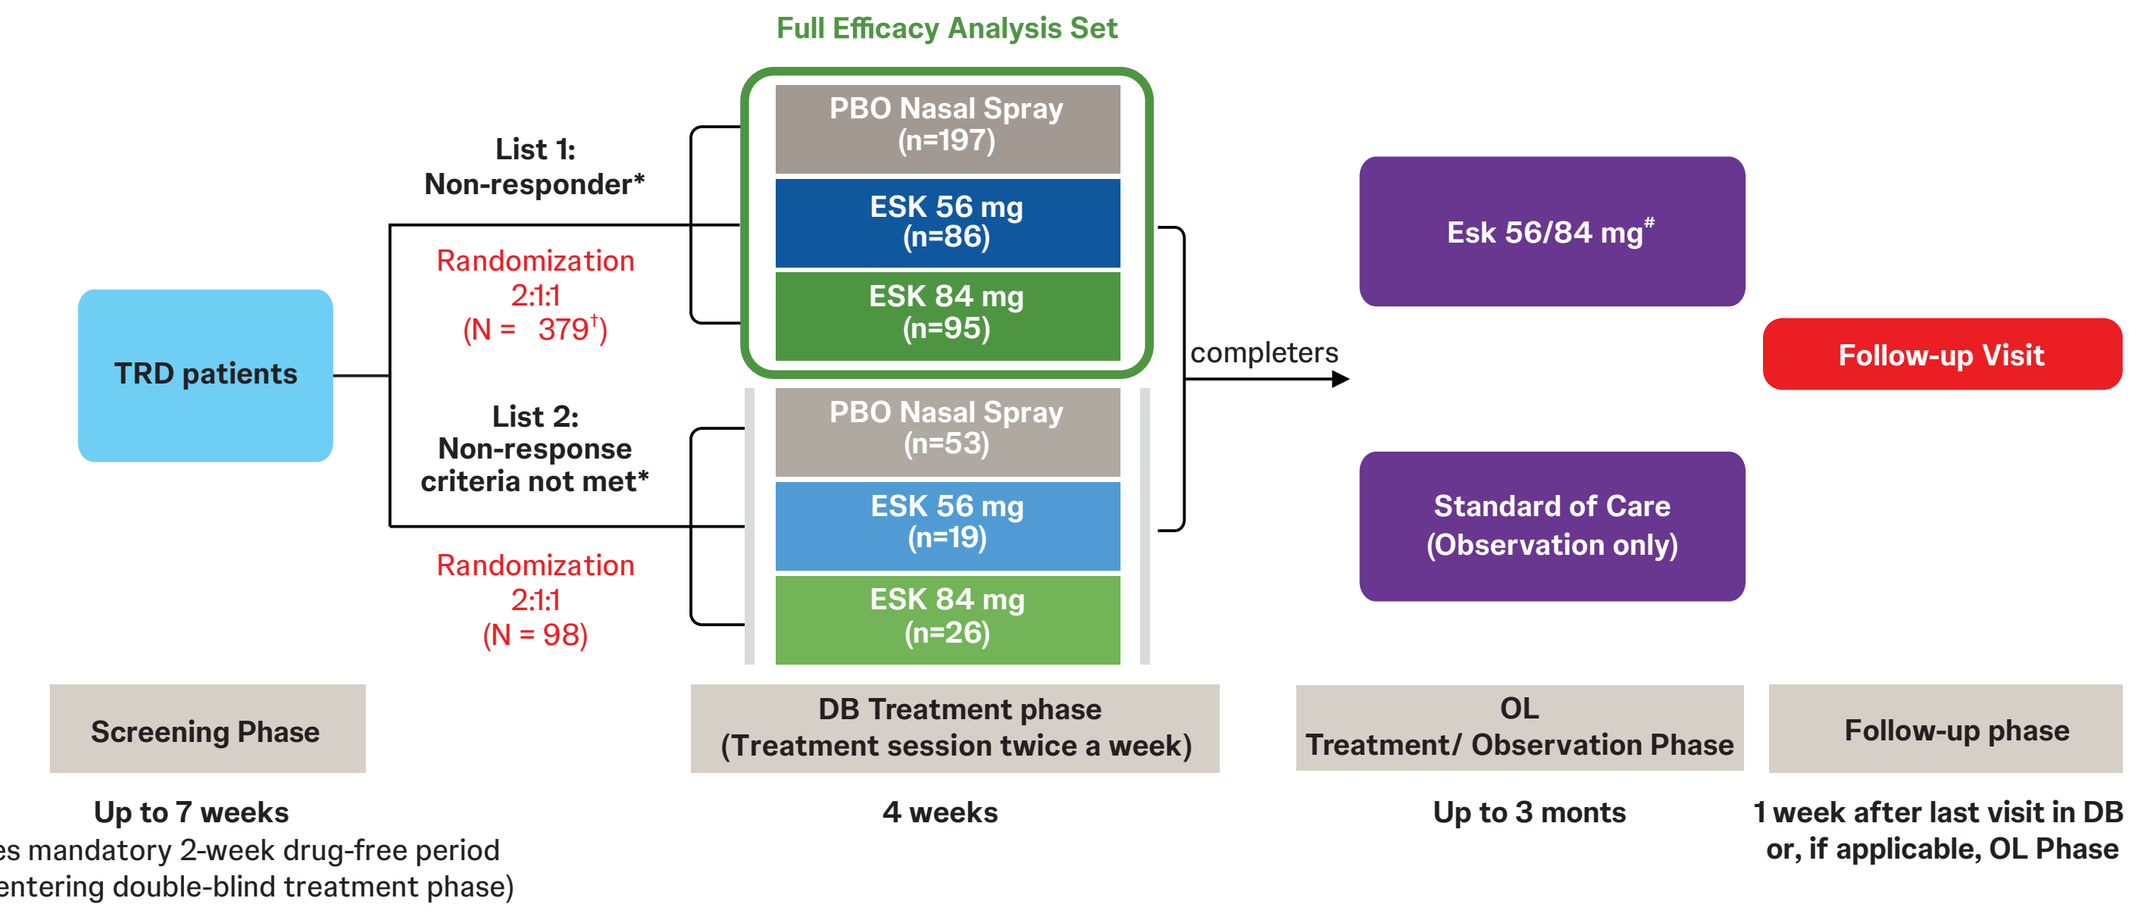
Full Efficacy Analysis Set
List 1: Non-responder*
Randomization 2:1:1 (N = 379†)
TRD patients
List 2: Non-response criteria not met*
Randomization 2:1:1 (N = 98)
PBO Nasal Spray (n=197)
ESK 56 mg (n=86)
ESK 84 mg (n=95)
PBO Nasal Spray (n=53)
ESK 56 mg (n=19)
ESK 84 mg (n=26)
completers
Esk 56/84 mg#
Standard of Care (Observation only)
Follow-up Visit
Screening Phase
DB Treatment phase (Treatment session twice a week)
OL Treatment/ Observation Phase
Follow-up phase
Up to 7 weeks (includes mandatory 2-week drug-free period prior to entering double-blind treatment phase)
4 weeks
Up to 3 monts
1 week after last visit in DB or, if applicable, OL Phase

*Non-response criteria (blinded to study sites): MADRS, total score of ≥ 28 at screening week 1, week 2 and day 1 (pre-randomization) and ≤ 25% improvement in the MADRS total score from screening week 1 to day 1 (pre-randomization). The non-responder criteria were designed to ensure the full efficacy analysis set included only those participants who met the severity criteria throughout screening without notable improvement in depressive symptoms. †One patient was not treated. #With or without standard of care. AD, antidepressant; DB, double-blind; ESK, esketamine; MADRS, Montgomery-Asberg Depression Rating Scale; OL, open-label; PBO, placebo; TRD, treatment-resistant depression.

Study design

- Randomized, double-blind (DB), PBO-controlled, multicenter study (NCT04599855) conducted in the United States, (**Fig. 1**).

Study evaluations

- **Primary efficacy endpoint**: Change in Montgomery–Åsberg Depression Rating Scale (MADRS) total score from baseline to Day 28.
- **Key secondary endpoint:** Change in MADRS total score from baseline to Day 2 (approximately 24 hours post first dose).
- **Safety:** Treatment-emergent adverse events (TEAEs) were monitored throughout the study.

Statistical analyses

- **Assessments**
 - Primary and key secondary endpoints were analyzed using a mixed-effects model with repeated measures and a predefined testing hierarchy to control multiplicity. The model included treatment group, analysis center, antidepressant treatment status (on- or off-treatment) at screening entry day, and day-by-treatment interaction as fixed terms, and the baseline MADRS total score as a covariate.
 - TEAEs were summarized descriptively by treatment group.
- **Analysis sets**
 - Full efficacy analysis set: All randomized participants meeting non-response criteria and who received ≥1 dose of DB study medication.
 - Safety analysis set: All randomized participants who received ≥1 dose of DB study medication.

TABLE 1. Demographics and baseline characteristics (full efficacy analysis set)

	PBO N=197	ESK 56 mg N=86	ESK 84 mg N=95	Total N=378
Age, mean (SD), years	45.2 (13.77)	46.5 (14.18)	44.8 (14.65)	45.4 (14.06)
Women, n (%)	119 (60.4)	51 (59.3)	61 (64.2)	231 (61.1)
Race, n (%)				
White	171 (86.8)	76 (88.4)	81 (85.3)	328 (86.8)
Black or African American	13 (6.6)	4 (4.7)	8 (8.4)	25 (6.6)
Asian	5 (2.5)	2 (2.3)	4 (4.2)	11 (2.9)
Other, multiple, unknown, or not reported	8 (4.1)	4 (4.7)	2 (2.1)	14 (3.7)
AD status at screening / entry, n (%)				
On-treatment	124 (62.9)	59 (68.6)	65 (68.4)	248 (65.6)
Off-treatment	73 (37.1)	27 (31.4)	30 (31.6)	130 (34.4)
Age when diagnosed with MDD, mean (SD), years	25.9 (11.43)	24.5 (10.54)	25.8 (10.73)	25.5 (11.04)
Duration of current depressive episode, mean (SD), weeks	289.0 (325.75)	419.8 (488.38)	406.4 (449.61)	348.3 (403.98)
Number of episodes since diagnosis, n (%)				
1	36 (18.3)	16 (18.6)	25 (26.3)	77 (20.4)
2	34 (17.3)	16 (18.6)	15 (15.8)	65 (17.2)
≥3	127 (64.5)	54 (62.8)	55 (57.9)	236 (62.4)
Baseline MADRS total score, mean (SD)	37.5 (4.90)	37.5 (5.23)	36.6 (4.48)	37.3 (4.88)
Baseline CGI-S score, mean (SD)	4.9 (0.61)	5.0 (0.60)	4.9 (0.65)	4.9 (0.62)
Baseline PHQ-9 total score, mean (SD)	19.8 (4.07)	20.7 (3.43)	19.9 (3.79)	20.0 (3.87)
IDS-C30 total score, mean (SD)	46.2 (7.21)	45.8 (7.00)	44.7 (6.90)	45.8 (7.10)
History of suicidal ideation in past 6/12 months, n (%)	105 (53.3)	38 (44.2)	52 (54.7)	195 (51.6)
Number of prior ADs with non-response, n (%)				
2	117 (59.4)	49 (57.0)	58 (61.1)	224 (59.3)
≥3	80 (40.6)	37 (43.0)	37 (38.9)	154 (40.7)

AD, antidepressant; BMI, body mass index; CGI-S, Clinical Global Impression – Severity; ESK, esketamine; IDS-C30, Inventory of Depressive Symptomatology-Clinician rated, 30-item; MADRS, Montgomery-Asberg Depression Rating Scale; MDD, major depressive disorder; PHQ-9, Patient Health Questionnaire 9 item; SD, standard deviation.

Results

- Total 477 participants were randomized:
 - 379 (79.5%) met the non-response criteria: 378 received study medication (ESK: 56 mg: 86; 84 mg: 95; PBO: 197) = full efficacy analysis set (Completers: 358/378, 94.7%), (**Fig. 2**).
 - 98 (20.5%) did not meet non-response criteria: received study medication and were included in the safety analysis set.

FIGURE 2. Participant disposition - DB phase (full efficacy analysis set)

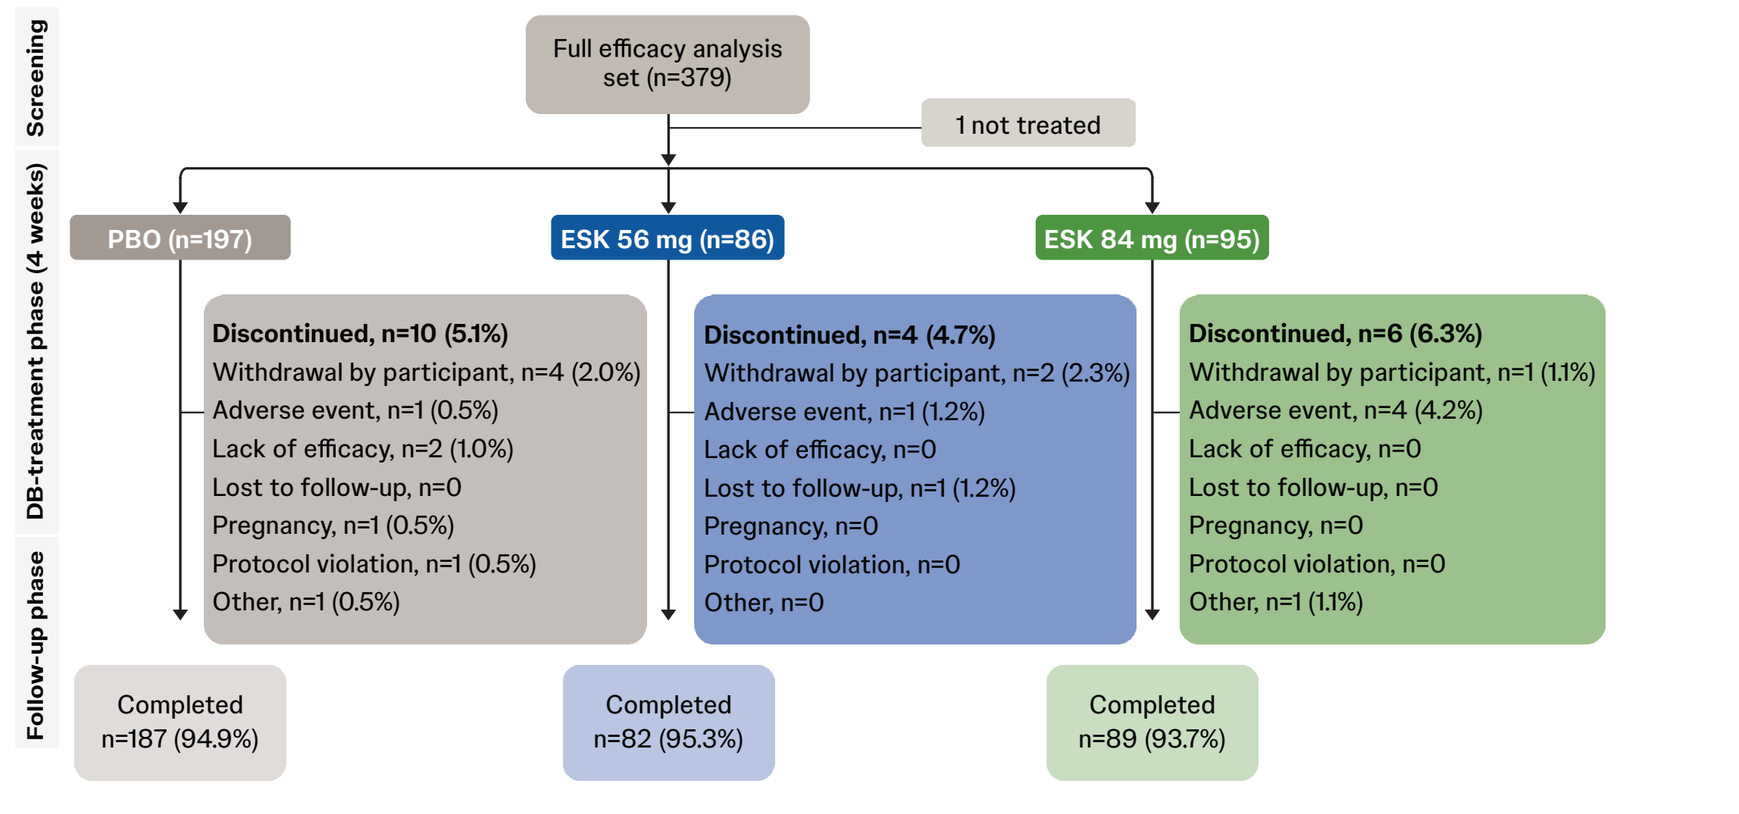
Screening
DB-treatment phase (4 weeks)
Follow-up phase
Full efficacy analysis set (n=379)
1 not treated
PBO (n=197)
ESK 56 mg (n=86)
ESK 84 mg (n=95)
Discontinued, n=10 (5.1%)
Withdrawal by participant, n=4 (2.0%)
Adverse event, n=1 (0.5%)
Lack of efficacy, n=2 (1.0%)
Lost to follow-up, n=0
Pregnancy, n=1 (0.5%)
Protocol violation, n=1 (0.5%)
Other, n=1 (0.5%)
Discontinued, n=4 (4.7%)
Withdrawal by participant, n=2 (2.3%)
Adverse event, n=1 (1.2%)
Lack of efficacy, n=0
Lost to follow-up, n=1 (1.2%)
Pregnancy, n=0
Protocol violation, n=0
Other, n=0
Discontinued, n=6 (6.3%)
Withdrawal by participant, n=1 (1.1%)
Adverse event, n=4 (4.2%)
Lack of efficacy, n=0
Lost to follow-up, n=0
Pregnancy, n=0
Protocol violation, n=0
Other, n=1 (1.1%)
Completed n=187 (94.9%)
Completed n=82 (95.3%)
Completed n=89 (93.7%)

DB, double-blind; ESK, esketamine; PBO, placebo.

- Demographic and baseline characteristics were comparable between the treatment groups (**Table 1**).
 - Most participants were women (231 [61.1%]), mean (SD) age was 45.4 (14.06) years, with 9.8% ≥65 years of age.
- At baseline, mean IDS-C30 score was 45.8; mean MADRS total score was 37.3.

Efficacy

- **Primary endpoint**
 - Mean MADRS total score decreased from baseline to Day 28, showing statistically significant and clinically meaningful improvement with individual doses of ESK vs PBO (2-sided p<0.001). The least-square mean difference (standard error [SE]) between ESK and PBO were: 56 mg, −5.1 (1.42) and 84 mg, −6.8 (1.38) (**Table 2, Fig. 3**).

TABLE 2. MADRS total score: change from baseline (full efficacy analysis set)

	PBO	ESK 56 mg	ESK 84 mg
Baseline (DB)			
N	197	86	95
Mean (SD)	37.5 (4.90)	37.5 (5.23)	36.6 (4.48)
Change from baseline to day 28			
N	185	82	89
Mean (SD)	−7.0 (10.07)	−12.7 (11.82)	−13.9 (11.89)
MMRM analysis			
Diff. of LS means (SE)		**−5.1 (1.42)**	**−6.8 (1.38)**
95% CI on diff		(−7.91; −2.33)	(−9.48; −4.07)
2-sided p-value		<0.001	<0.001
Change from baseline to day 2			
N	195	84	93
Mean (SD)	−9.7 (10.27)	−13.9 (10.15)	−13.0 (9.68)
MMRM analysis			
Diff. of LS means (SE)		**−3.8 (1.29)**	**−3.4 (1.24)**
95% CI on diff		(−6.29; −1.22)	(−5.89; −1.00)
2-sided p-value		0.004	0.006

Note: MADRS total score ranges from 0 to 60; a higher score indicates a more severe condition. Note: Negative change in score indicates improvement. CI, confidence interval; DB, double-blind; ESK, esketamine; LS, least square; MADRS, Montgomery-Asberg Depression Rating Scale; MMRM, mixed model for repeated measures; PBO, placebo; SD, standard deviation; SE, standard error.

FIGURE 3. MADRS total score: LS mean change (± SE) over time in the DB phase - MMRM observed case (full efficacy analysis set)

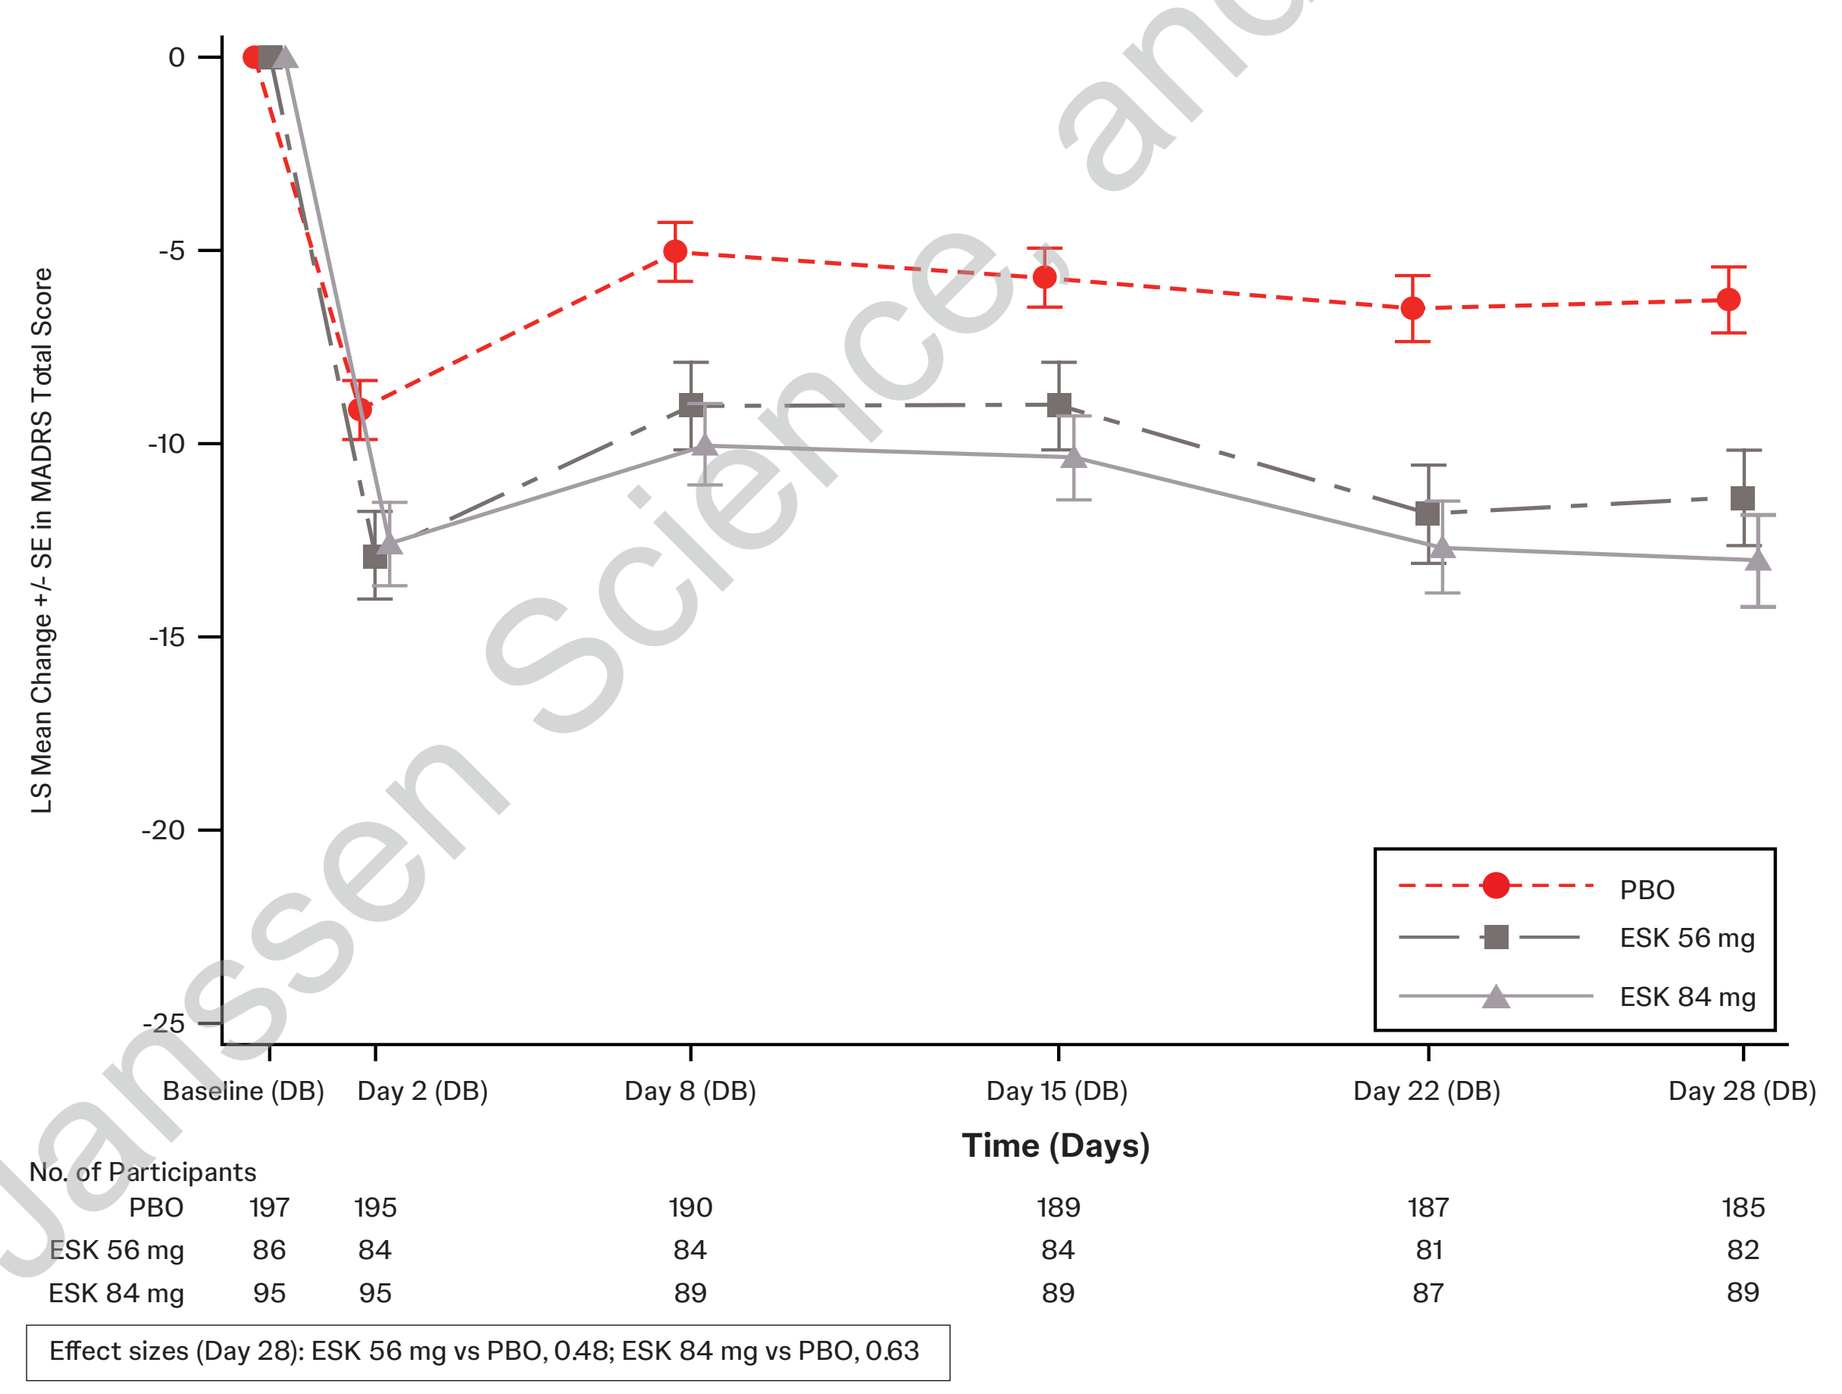
LS Mean Change +/- SE in MADRS Total Score
0
-5
-10
-15
-20
-25
PBO
ESK 56 mg
ESK 84 mg
Baseline (DB)
Day 2 (DB)
Day 8 (DB)
Day 15 (DB)
Day 22 (DB)
Day 28 (DB)
Time (Days)
No. of Participants
PBO 197 195 190 189 187 185
ESK 56 mg 86 84 84 84 81 82
ESK 84 mg 95 95 89 89 87 89
Effect sizes (Day 28): ESK 56 mg vs PBO, 0.48; ESK 84 mg vs PBO, 0.63

DB, double-blind; ESK, esketamine; LS, least squares; MADRS, Montgomery-Asberg Depression Rating Scale; MMRM, mixed model for repeated measures; PBO, placebo; SE, standard error.

- **Key secondary endpoint**
 - Significantly greater improvement was noted in the ESK 56 mg group (2-sided p=0.004) and ESK 84 mg group (2-sided p=0.006) vs PBO. On Day 2, LS mean difference (SE) between ESK, and PBO was −3.8 (1.29) for 56 mg and −3.4 (1.24) for 84 mg (**Table 2**).
- **Other secondary endpoints**
 - Response rates based on MADRS total score
 - Higher response rates (≥50% reduction from baseline in MADRS total score) were observed in both ESK groups vs PBO at all DB timepoints (**Fig. 4**).
 - Response rate at Day 28: ESK 56 mg: 30.5%; ESK 84 mg: 29.2%; PBO: 15.1%.

FIGURE 4. MADRS total score: Participants achieving response (≥50% improvement) over time in the DB Phase (full efficacy analysis set)

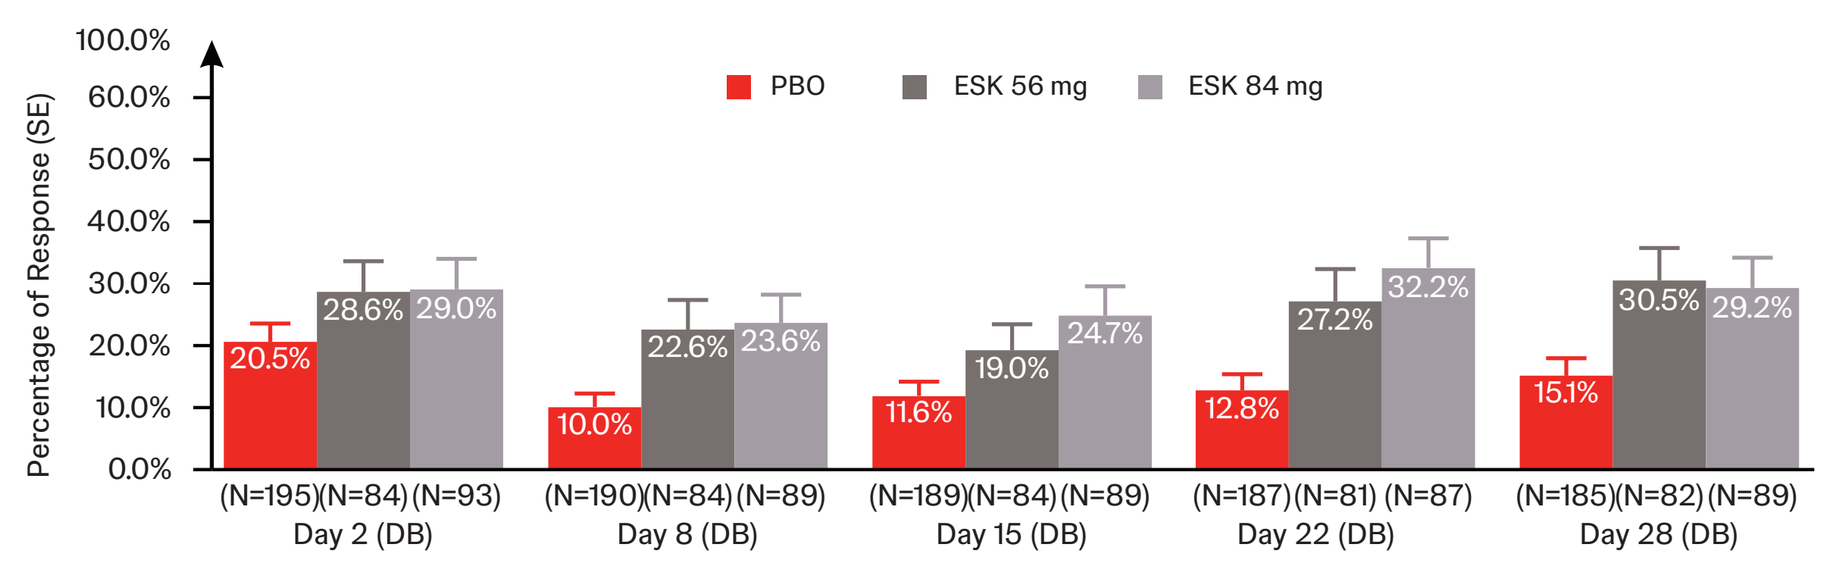
Percentage of Response (SE)
100.0%
60.0%
50.0%
40.0%
30.0%
20.0%
10.0%
0.0%
PBO
ESK 56 mg
ESK 84 mg
20.5%
28.6%
29.0%
10.0%
22.6%
23.6%
11.6%
19.0%
24.7%
12.8%
27.2%
32.2%
15.1%
30.5%
29.2%
(N=195)(N=84)(N=93) Day 2 (DB)
(N=190)(N=84)(N=89) Day 8 (DB)
(N=189)(N=84)(N=89) Day 15 (DB)
(N=187)(N=81) (N=87) Day 22 (DB)
(N=185)(N=82)(N=89) Day 28 (DB)

DB, double-blind; ESK, esketamine; MADRS, Montgomery-Asberg Depression Rating Scale; PBO, placebo; SE, standard error. Note: Percentages are calculated with N at each time as the denominator.

Remission rates based on MADRS total score

- Higher remission rates (MADRS ≤10 and MADRS ≤12) were observed in both ESK vs PBO groups at all DB timepoints (**Fig. 5**).
 Remission rates at Day 28:
 MADRS ≤10: ESK 56 mg: 14.6%; ESK 84 mg: 21.3%; PBO: 6.5%.
 MADRS ≤12: ESK 56 mg: 18.3%; ESK 84 mg: 22.5%; PBO: 7.6%

Figure 5: MADRS total score: Participants achieving remission (MADRS ≤10) over time in the DB Phase (full efficacy analysis set)

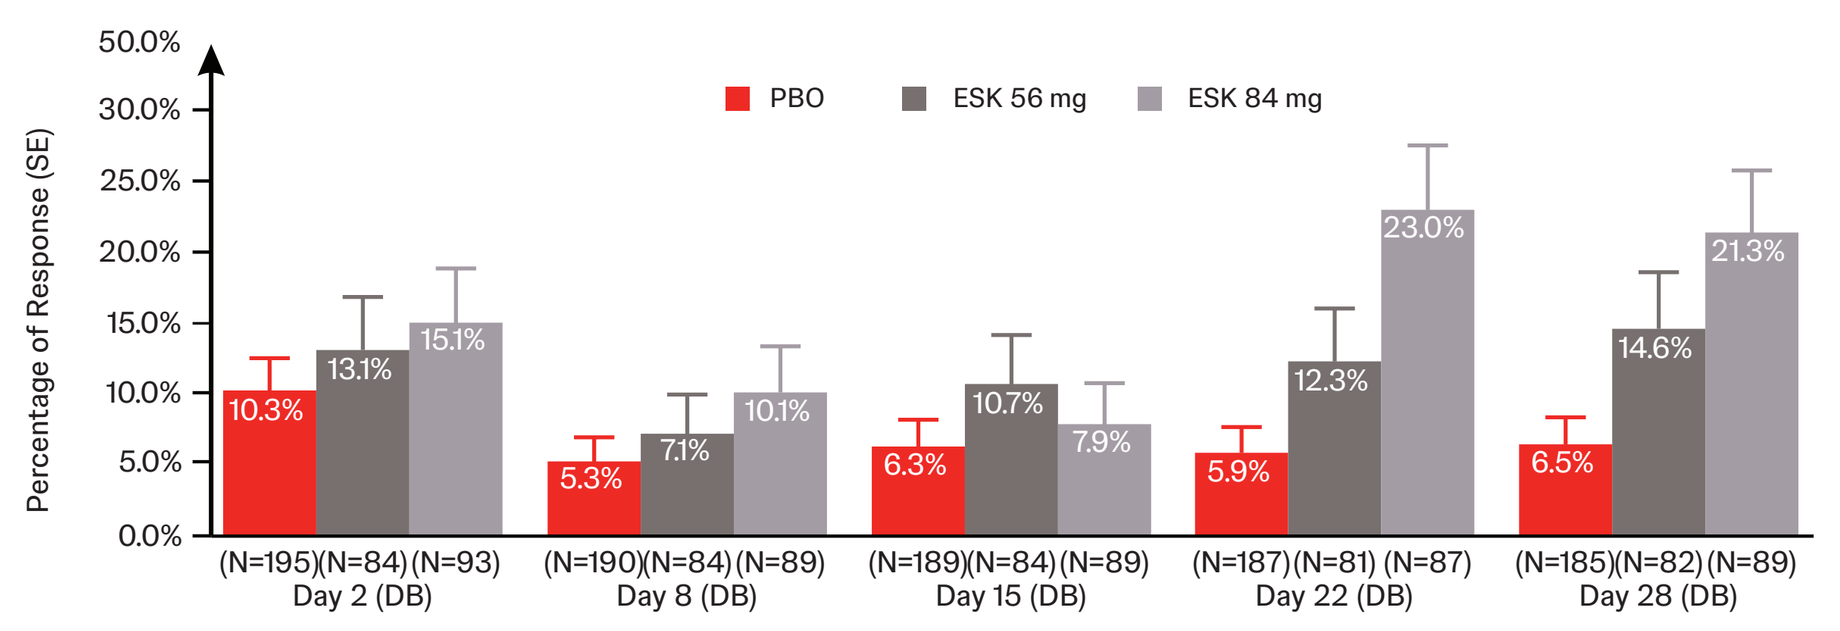
Percentage of Response (SE)
50.0%
30.0%
25.0%
20.0%
15.0%
10.0%
5.0%
0.0%
PBO
ESK 56 mg
ESK 84 mg
10.3%
13.1%
15.1%
5.3%
7.1%
10.1%
6.3%
10.7%
7.9%
5.9%
12.3%
23.0%
6.5%
14.6%
21.3%
(N=195)(N=84)(N=93) Day 2 (DB)
(N=190)(N=84)(N=89) Day 8 (DB)
(N=189)(N=84)(N=89) Day 15 (DB)
(N=187)(N=81) (N=87) Day 22 (DB)
(N=185)(N=82)(N=89) Day 28 (DB)

DB, double-blind; ESK, esketamine; MADRS, Montgomery-Asberg Depression Rating Scale; PBO, placebo; SE, standard error. Note: Percentages are calculated with N at each time as the denominator.

Safety

- ≥1TEAE during DB phase: combined ESK: 73.9% (ESK 56 mg: 72.4% and ESK 84 mg: 75.2%); PBO: 49.2%; the majority of TEAEs were transient.
- The most common (>10%) TEAEs during DB phase in combined ESK group vs PBO were nausea, dissociation, dizziness, and headache (**Table 3**).
- Serious TEAEs were reported in 6 participants in the DB phase: ESK 56 mg: ankle fracture (n=1); ESK 84 mg: ophthalmic migraine and suicide attempt (n=1 each); PBO: self-injurious ideation, suicidal ideation, and acute myocardial infarction (n=1 each). None of these (except acute myocardial infarction: PBO), were considered related to the study medication.
- No deaths were reported in either the DB or open-label phase.

Table 3. Most frequently reported TEAEs* in the DB treatment phase (safety analysis set)

TEAE	PBO, n (%) N=250	ESK, n (%) 56 mg N=105	ESK, n (%) 84 mg N=121	ESK, n (%) Combined N=226
Nausea	21 (8.4)	24 (22.9)	32 (26.4)	56 (24.8)
Dissociation	7 (2.8)	23 (21.9)	32 (26.4)	55 (24.3)
Dizziness	18 (7.2)	22 (21.0)	27 (22.3)	49 (21.7)
Headache	22 (8.8)	19 (18.1)	24 (19.8)	43 (19.0)
Feeling drunk	2 (0.8)	8 (7.6)	8 (6.6)	16 (7.1)
Anxiety	3 (1.2)	5 (4.8)	10 (8.3)	15 (6.6)
Fatigue	11 (4.4)	8 (7.6)	7 (5.8)	15 (6.6)
Vomiting	1 (0.4)	5 (4.8)	10 (8.3)	15 (6.6)
Insomnia	9 (3.6)	6 (5.7)	5 (4.1)	11 (4.9)
Somnolence	4 (1.6)	6 (5.7)	3 (2.5)	9 (4.0)

*Incidence ≥5% in either treatment group. Note: TEAEs listed in decreasing order based on incidence within the combined esketamine group, and in alphabetical order for events with the same incidence. DB, double-blind; ESK, esketamine; PBO, placebo; TEAE, treatment-emergent adverse event.

Conclusions

- The study met primary and key secondary efficacy objectives.
- In patients with TRD, esketamine (56 mg and 84 mg) as monotherapy showed statistically significant and clinically meaningful improvement in depressive symptoms compared to placebo after 4 weeks of treatment (primary endpoint), and as early as Day 2 (approximately 24 hours post first dose: key secondary endpoint).
- Through 4 weeks of double-blind treatment phase, both esketamine (56 mg and 84 mg) doses showed higher response rates and remission rates compared to placebo (other secondary endpoints).
- The safety profile of esketamine as monotherapy was consistent with the well-established safety profile of esketamine from prior adjunctive treatment studies.
- These results provide important data demonstrating monotherapy regimens for patients with TRD.

Acknowledgements

The authors thank the participants and investigators for their participation in the study. Kalpana Tilekar, PhD (SIRO Clinpharm Pvt. Ltd., India) provided writing assistance and Ellen Baum, PhD (Janssen Global Services, LLC) provided additional editorial support. Mugdha Rokade (SIRO Clinpharm Pvt. Ltd., India) provided graphic designing support.

Disclosures

All authors are employees of Janssen Research & Development, LLC and hold stocks or stock options in Johnson & Johnson.

Previous Presentation

The data in this poster were previously presented at the American Psychiatric Nurses Association (APNA) 38th Annual Conference; Oct 9-12, 2024; Louisville, Kentucky.

Novel Pathways in Depression

Scan the QR code

The QR code is intended to provide scientific information for individual reference, and the information should not be altered or reproduced in any way.

Supported by Janssen Scientific Affairs, LLC, a Johnson & Johnson company

Presented at NEI Congress; November 7-10, 2024; Colorado Springs, Colorado

References

1. APA CLINICAL PRACTICE GUIDELINE for the Treatment of Depression Across Three Age Cohorts. https://www.apa.org/depression-guideline/guideline.pdf. Accessed 22 August 2024. 2. Popova V, et al. *AM J Psychiatry*. 2019; 176(6):428–438. 3. Reif A, et al. *N Engl J Med*. 2023; 389(14):1298–1309.

This material is distributed for scientific purposes on Janssen Science, and is not for promotional use