Total Cost per Remitter of Lumateperone Versus Cariprazine for the Treatment of Major Depressive Disorder in the United States

Eric Wu[1], Michael K Xu[1], Jack Eales[1], Hekun Chen[1], Zheng-Yi Zhou[1], Madhav Namjoshi[2]

[1]Analysis Group Inc., Boston, MA, USA [2]Johnson & Johnson Innovative Medicine, Raritan, NJ, USA

Poster # 79

Background

- The economic burden of major depressive disorder (MDD) in the United States (US) is substantial, having been estimated at $326.2 billion in 2018 (2020 US dollars [USD]), a 37.9% increase from 2010 due to growing workplace costs as the affected population has trended younger[1]
- Remission is the ultimate goal of MDD treatment. However, approximately 30% of patients fail to achieve remission after multiple sequential treatments.[2] For patients with inadequate response to antidepressant therapy, atypical antipsychotics may be used as adjunctive treatment
- Cariprazine (approved December 2022) and lumateperone (approved November 2025) are the two most recent US Food and Drug Administration (FDA) approvals of atypical antipsychotics for adjunctive treatment of MDD – the first since brexpiprazole in 2015. Both also report 26-week open-label extension (OLE) data in adjunctive MDD, making them directly comparable on longer-term remission
- No comparative economic analysis between these two agents has yet been published. This study was conducted to help inform reimbursement decisions for payers by incorporating both clinical and economic outcomes in a cost per remitter analysis

Objective

- This study aimed to evaluate the total cost per remitter and cost per responder for lumateperone versus cariprazine as adjunctive treatment for adult patients with MDD over a 6-month time horizon from a US commercial payer perspective

Methods

- An economic model was built in Microsoft Excel 365 to calculate the cost per remitter and cost per responder of patients receiving lumateperone versus cariprazine as adjunctive treatment to antidepressants for adult patients with MDD during the first 6 months of treatment, from a US commercial payer perspective

Costs

- The costs associated with drug acquisition, monitoring, adverse event (AE), and remission- or response-related medical costs were considered in the model. Cost inputs and their respective sources are listed in **Table 1**
 - Drug acquisition costs were derived based on the unit drug costs (wholesale acquisition cost from Merative Micromedex RED BOOK), dosing schedules, and time on treatment. Time on treatment was calculated assuming an exponential distribution, calculated based on the reported mean duration of exposure from each comparator's OLE studies
 - Monitoring costs included costs for recommended regular treatment-related monitoring procedures per prescribing information, including physician visit, blood glucose testing, and lipid profile testing; frequencies were informed by the treatment guidelines for bipolar disorder, as MDD-specific frequencies were not listed in relevant prescribing information. Unit costs were obtained from Centers for Medicare & Medicaid Services (CMS) sources
 - AE costs were calculated based on AE rates for each treatment as reported in the respective OLE studies, and unit costs per AE based on literature sources. AEs considered in the model were extrapyramidal symptoms (EPS) and weight gain
 - Costs associated with remission or response were taken from a study examining costs associated with MDD severity in patients who had failed prior medication treatment.[3] Costs reported based on Patient Health Questionnaire-9 (PHQ-9) severity categories were mapped to Montgomery-Åsberg Depression Rating Scale (MADRS) total score categories[4]

Effectiveness

- 6-month remission and response rates for lumateperone and cariprazine were obtained from their respective 26-week phase 3 OLE studies (Study 503 [Durgam et al. 2026] for lumateperone; Vieta et al. 2019 for cariprazine) and are presented in **Table 1**
 - Study 503 evaluated the long-term safety and effectiveness of adjunctive lumateperone in patients who completed Study 501 or 502. Results showed improvements in depressive symptoms were maintained throughout the OLE period, with 65.4% of patients achieving remission and 79.5% of patients achieving clinically defined response at end of open-label treatment[5]
 - Both 26-week OLE studies enrolled patients rolling over from double-blind, placebo-controlled lead-in trials of adjunctive therapy for MDD (Studies 501 and 502 for lumateperone; a negative 8-week trial for cariprazine, whose rollover patients entered the OLE after a 2-week washout on antidepressants alone, and which also enrolled cariprazine-naïve patients)[5,6]
 - In both studies, remission was defined as a MADRS total score of ≤10; and response was defined as a ≥50% reduction from baseline in the MADRS total score
 - For cariprazine, efficacy outcomes were reported using two approaches for imputing missing data: (1) last observation carried forward (LOCF), and (2) observed cases (OC).[6] As the reported OC remission and response rates were higher than those under LOCF, the model used the OC estimates in the base case, with LOCF estimates explored in scenario analysis

Table 1. Model inputs

Cost input	Lumateperone	Cariprazine	Source
Unit drug acquisition cost per dose, 2026 USD	$59.85 (42 mg)	$53.16 (1.5, 3.0, and 4.5 mg)	Merative Micromedex RED BOOK[7]
Mean duration of exposure, months	5.3	4.4	Durgam et al. 2026[5]; Vieta et al. 2019[6]
Monitoring costs, 2026 USD	$235	$215	CMS Laboratory Fee Schedule[8]; CMS Physician Fee Schedule[9] Dodd et al. 2018[10]; CAPLYTA Prescribing information[11]; VRAYLAR Prescribing information[12]
Adverse event costs, 2026 USD	Extrapyramidal symptoms: $6,005 Weight gain: $9,134		Kadakia et al. 2022[13]; Tkacz et al. 2026[14]
Proportion of patients experiencing extrapyramidal symptoms (%)	3.8%	28.4%	Durgam et al. 2026[5]; Vieta et al. 2019[6]
Proportion of patients experiencing weight gain (%)	8.5%	19.4%	
Remission- or response-related medical costs, 2026 USD	Remission: $7,412 Response without remission: $12,026 Non-response / non-remission: $14,970		Culpepper et al. 2024[3]; Kim et al. 2024[4]
Efficacy inputs	**Lumateperone**	**Cariprazine**	**Source**
Remission rates	65.4%	OC: 53.3%; LOCF: 45.8%	Lumateperone: Durgam et al. 2026[5]; Cariprazine: Vieta et al. 2019[6]
Response rates	79.5%	OC: 43.3%; LOCF: 37.2%	

Abbreviations: : CMS, Centers for Medicare & Medicaid Services; LOCF, last observation carried forward; OC, observed cases; USD, US dollars.

Key Findings

- Over a 6-month time horizon, total cost of care was $12,733 for lumateperone and $14,610 for cariprazine (**Figure 1**)
 - Drug acquisition costs were higher for lumateperone than cariprazine ($6,689 vs. $5,446), driven by longer mean treatment exposure. However, these were offset by lower AE costs ($1,004 vs. $3,479) and lower remission- or response-related medical costs ($4,806 vs. $5,470), due to the higher remission rate for lumateperone. Monitoring costs were similar between treatments (lumateperone: $235 vs. cariprazine: $215)
- Lumateperone had a lower total cost per remitter ($19,470) than cariprazine ($27,393) over the modeled time horizon
 - This difference was driven by a combination of both lower total costs and higher remission rate for lumateperone versus cariprazine (65.4% vs. 53.3%)
 - Lumateperone also had a lower total cost per responder than cariprazine ($16,017 vs. $33,715), driven by lower total costs and a higher response rate for lumateperone versus cariprazine (79.5% vs. 43.3%)
- In a scenario analysis using efficacy outcomes imputed using LOCF for cariprazine, results were consistent with the base case. Lower imputed cariprazine remission and response rates under LOCF led to more favorable results for lumateperone

Table 2. Results Comparing Lumateperone and Cariprazine over a 6-month Time Horizon

Cost Components (2026 USD)	Lumateperone	Cariprazine	Difference
Drug acquisition costs	$6,689	$5,446	$1,243
Monitoring costs	$235	$215	$20
Adverse event costs	$1,004	$3,479	-$2,476
Remission- or response-related medical costs	$4,806	$5,470	-$664
Total costs per treated patient	**$12,733**	**$14,610**	**-$1,876**
Remission rate	65.4%	53.3%	-
Response rate	79.5%	43.3%	-
Total costs per remitter	**$19,470**	**$27,393**	**-$7,923**
Total costs per responder	**$16,017**	**$33,715**	**-$17,698**

Abbreviations: OC, observed cases; USD, US dollars.
Note: Results presented using OC values for cariprazine remission and response rates.

Figure 1. Total Cost of Care of Lumateperone and Cariprazine over a 6-month Time Horizon

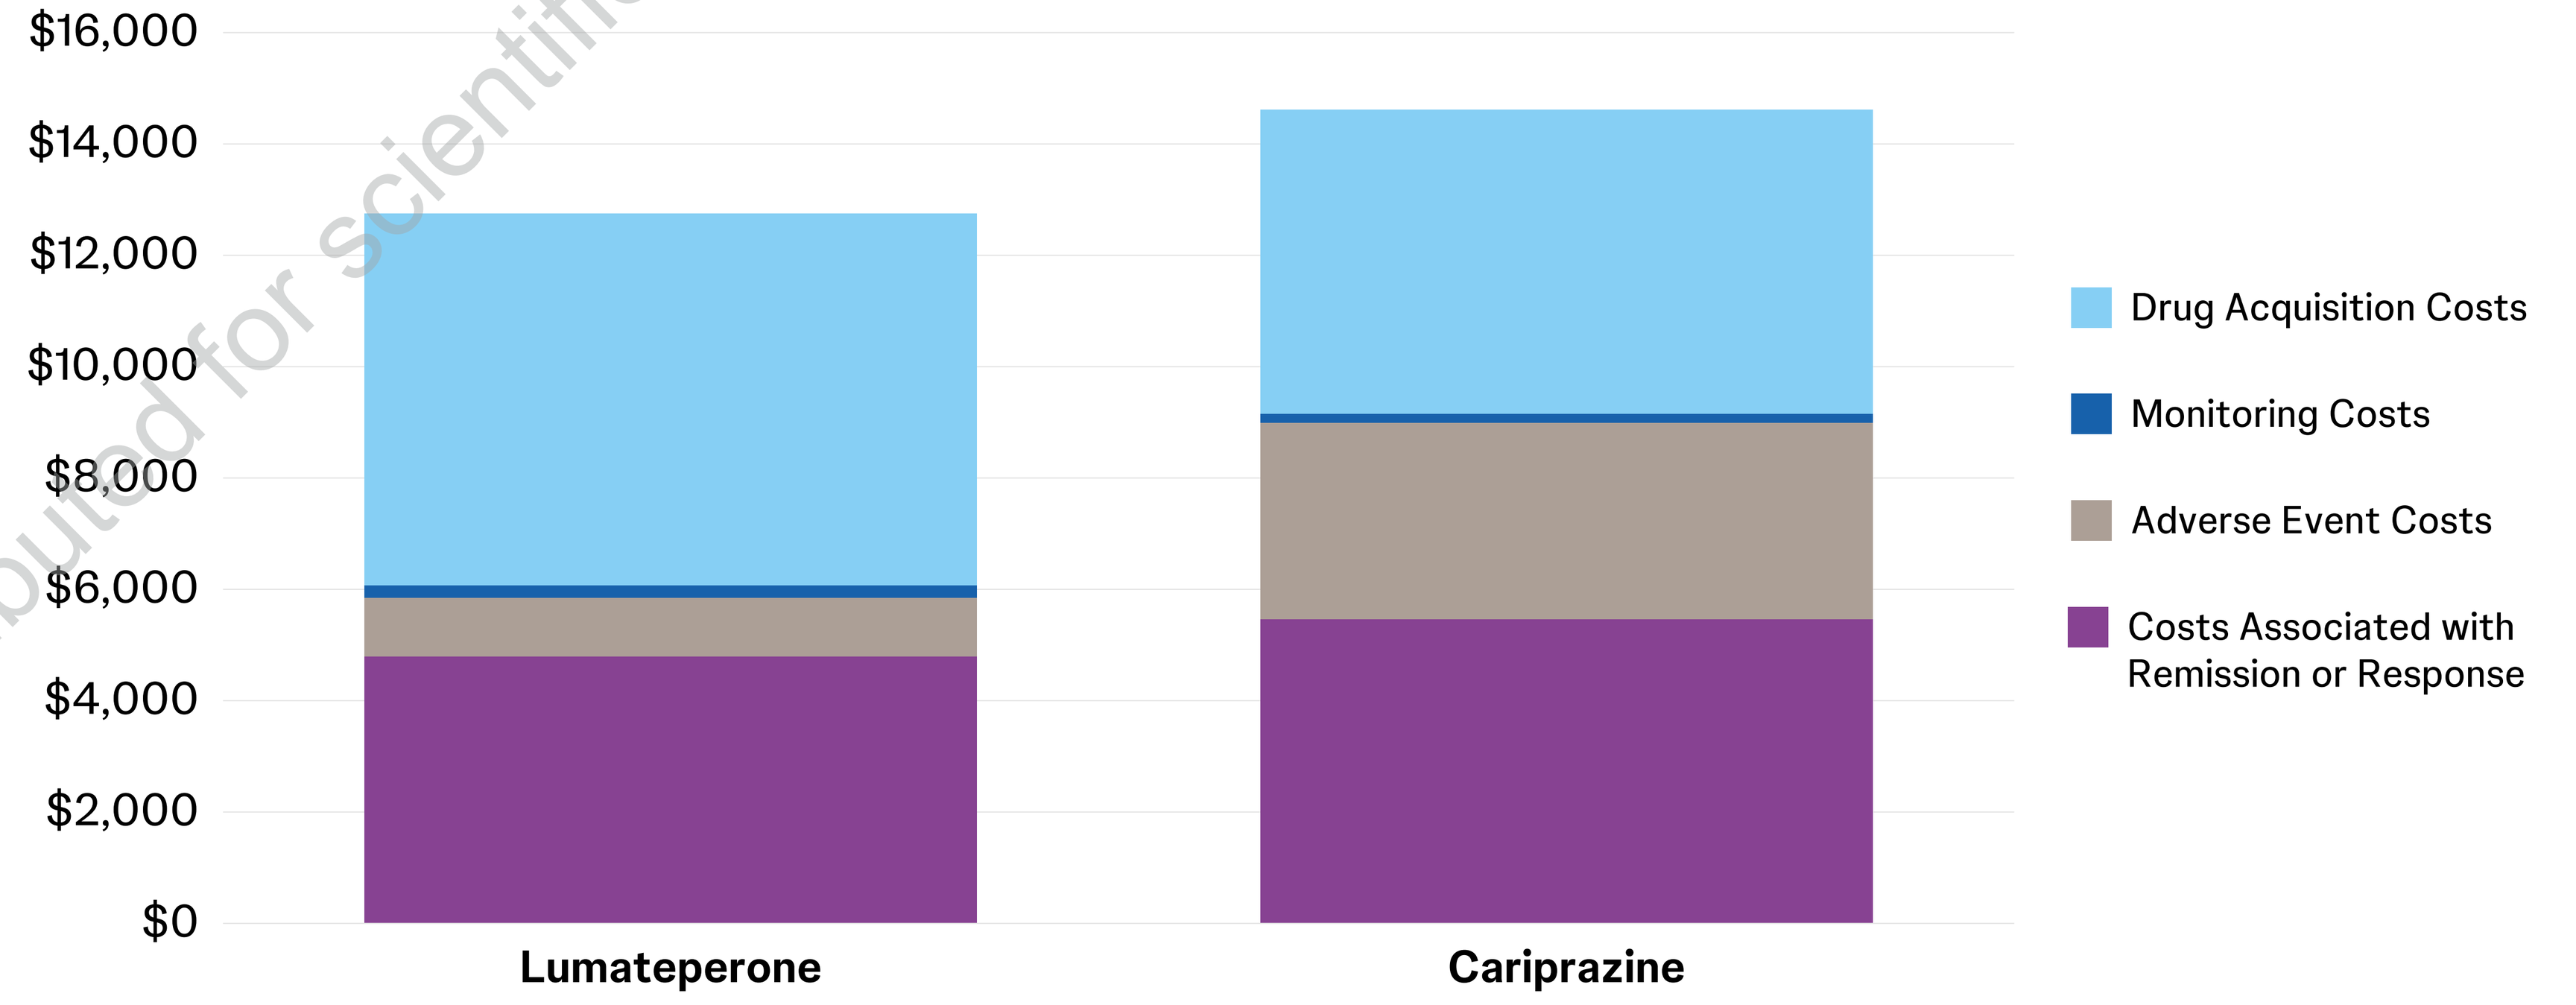
$16,000
$14,000
$12,000
$10,000
$8,000
$6,000
$4,000
$2,000
$0
Lumateperone
Cariprazine
Drug Acquisition Costs
Monitoring Costs
Adverse Event Costs
Costs Associated with Remission or Response

Note: Results presented using OC values for cariprazine remission and response rates.

- Findings were robust across sensitivity analyses, with results most sensitive to mean duration of exposure, AE costs, and costs associated with non-response/remission; all results continued to favor lumateperone over cariprazine (**Figures 2 and 3**)

Figure 2. Sensitivity Analysis Results: Difference in Total Cost per Remitter – Lumateperone vs. Cariprazine

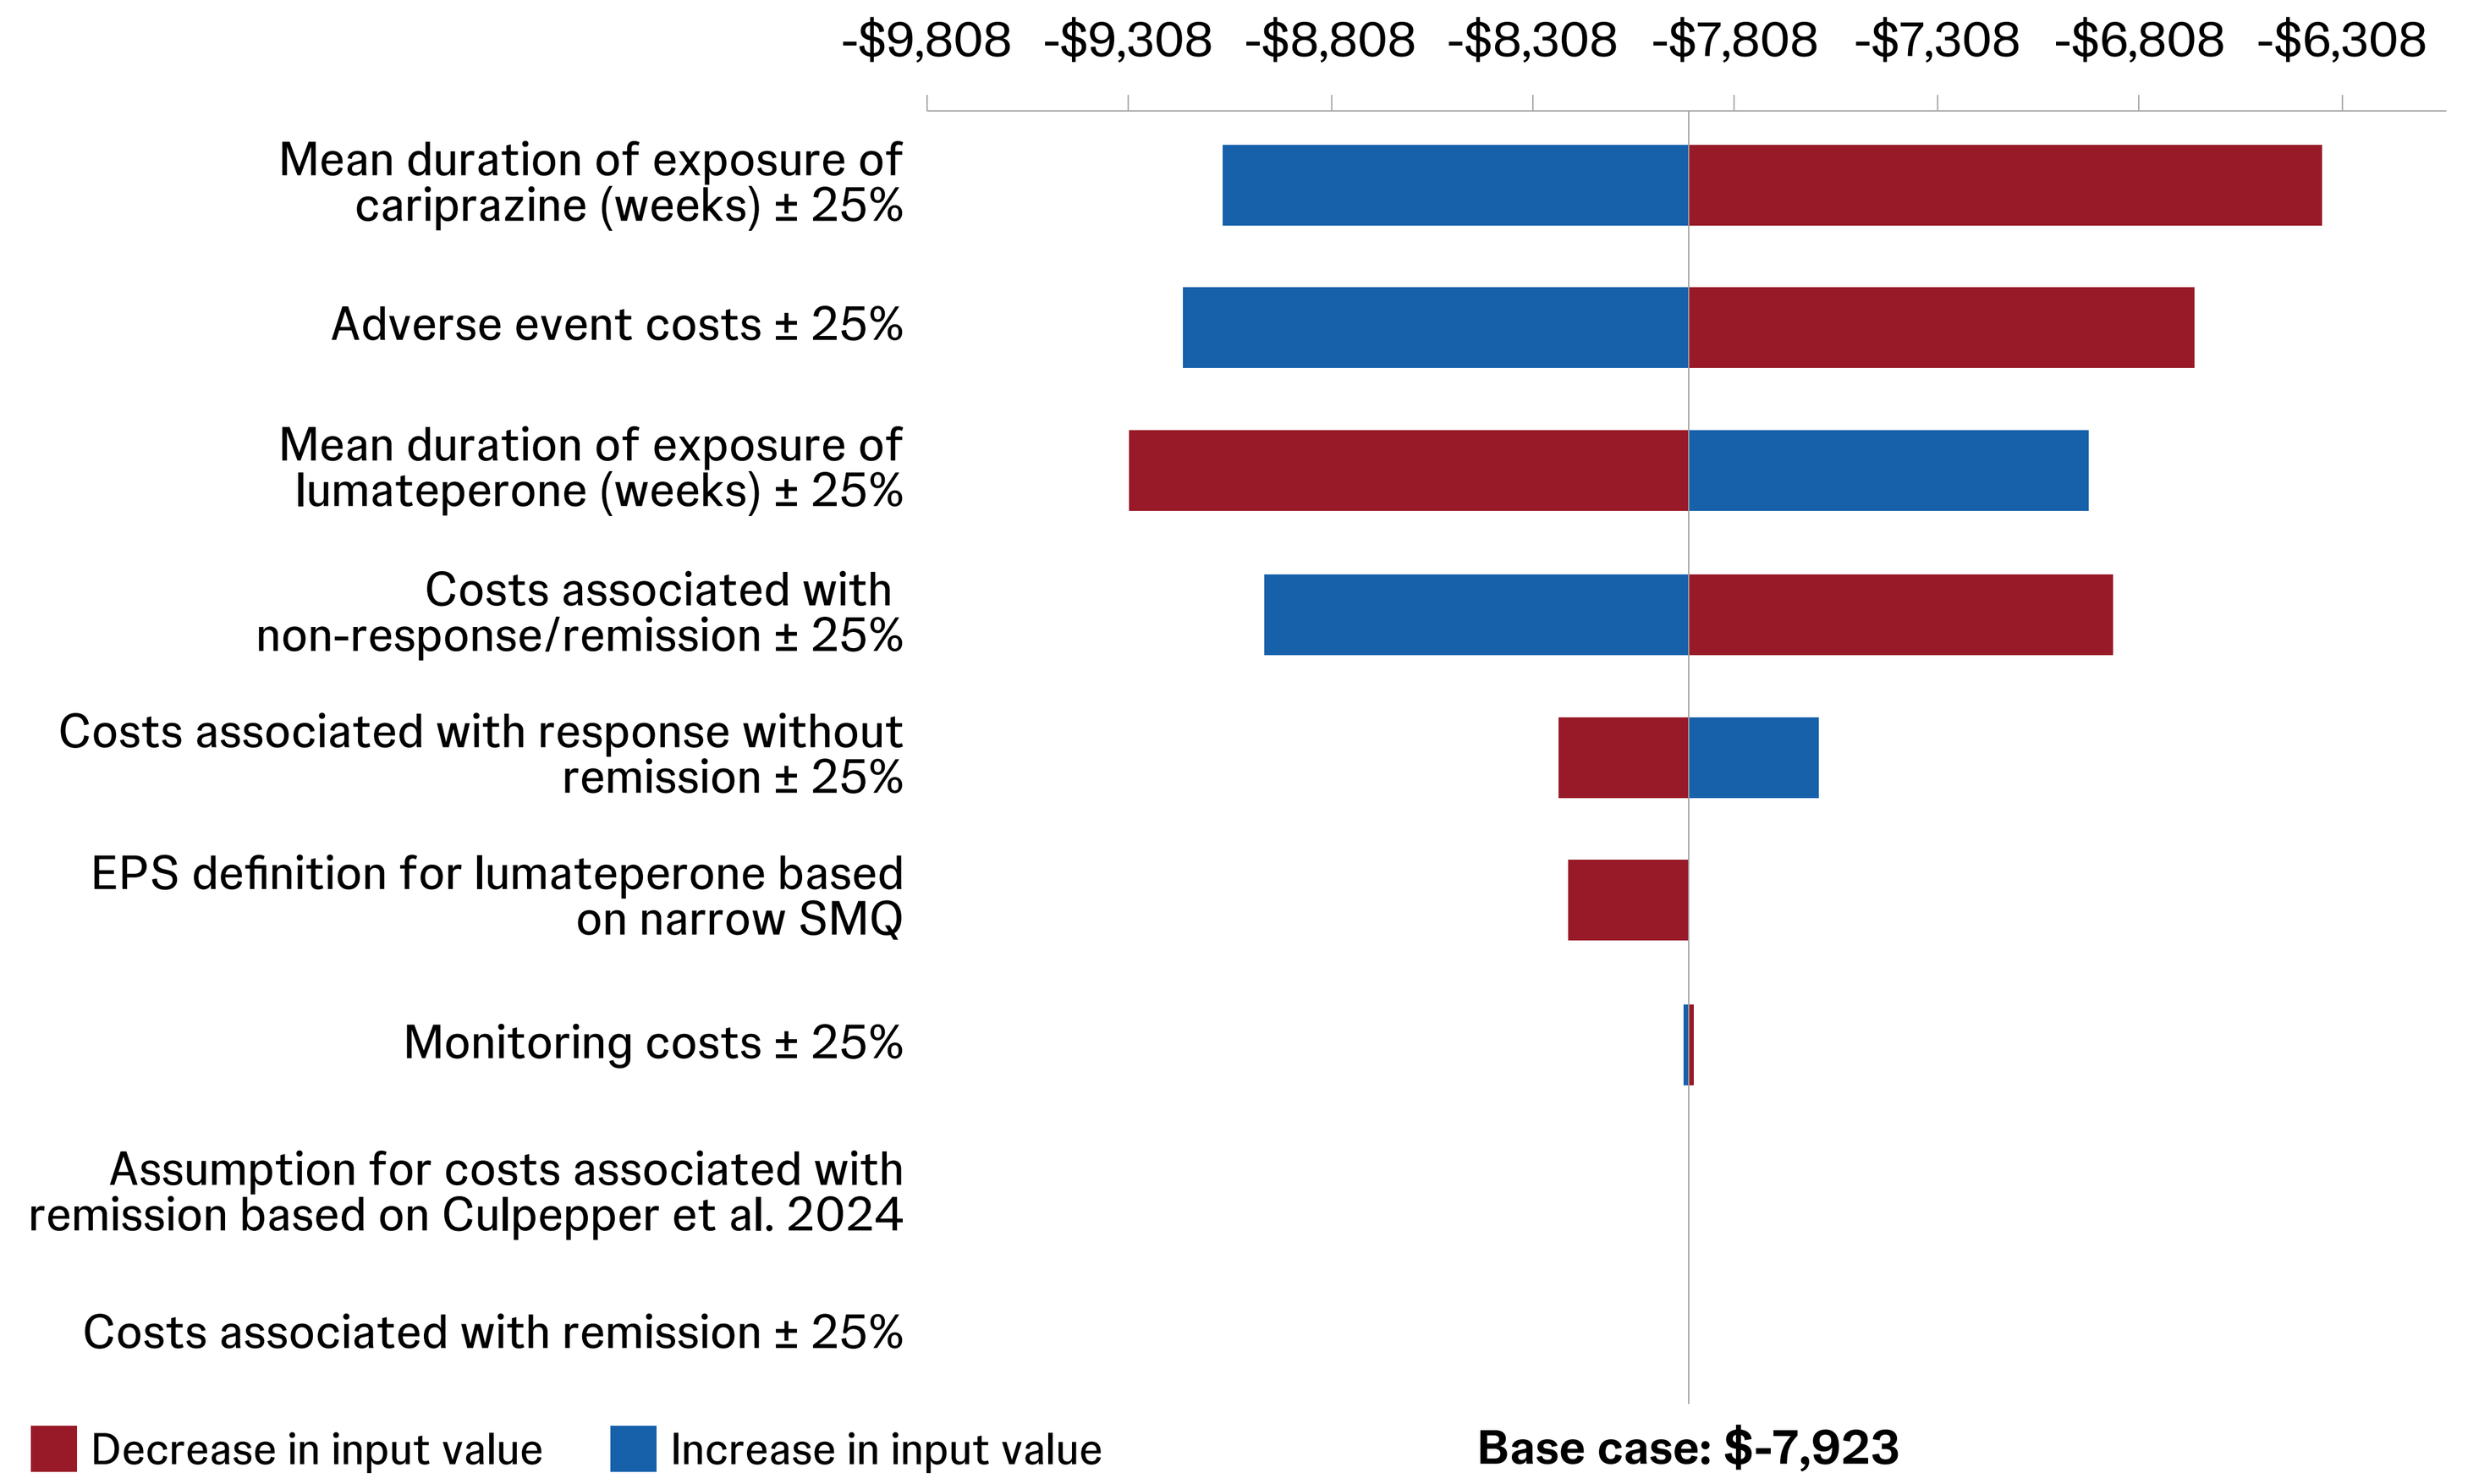
-$9,808
-$9,308
-$8,808
-$8,308
-$7,808
-$7,308
-$6,808
-$6,308
Mean duration of exposure of cariprazine (weeks) ± 25%
Adverse event costs ± 25%
Mean duration of exposure of lumateperone (weeks) ± 25%
Costs associated with non-response/remission ± 25%
Costs associated with response without remission ± 25%
EPS definition for lumateperone based on narrow SMQ
Monitoring costs ± 25%
Assumption for costs associated with remission based on Culpepper et al. 2024
Costs associated with remission ± 25%
Decrease in input value
Increase in input value
Base case: $-7,923

Abbreviations: EPS, extrapyramidal symptoms; SMQ, Standardised MedDRA Query.

Figure 3. Sensitivity Analysis Results: Difference in Total Cost per Responder – Lumateperone vs. Cariprazine

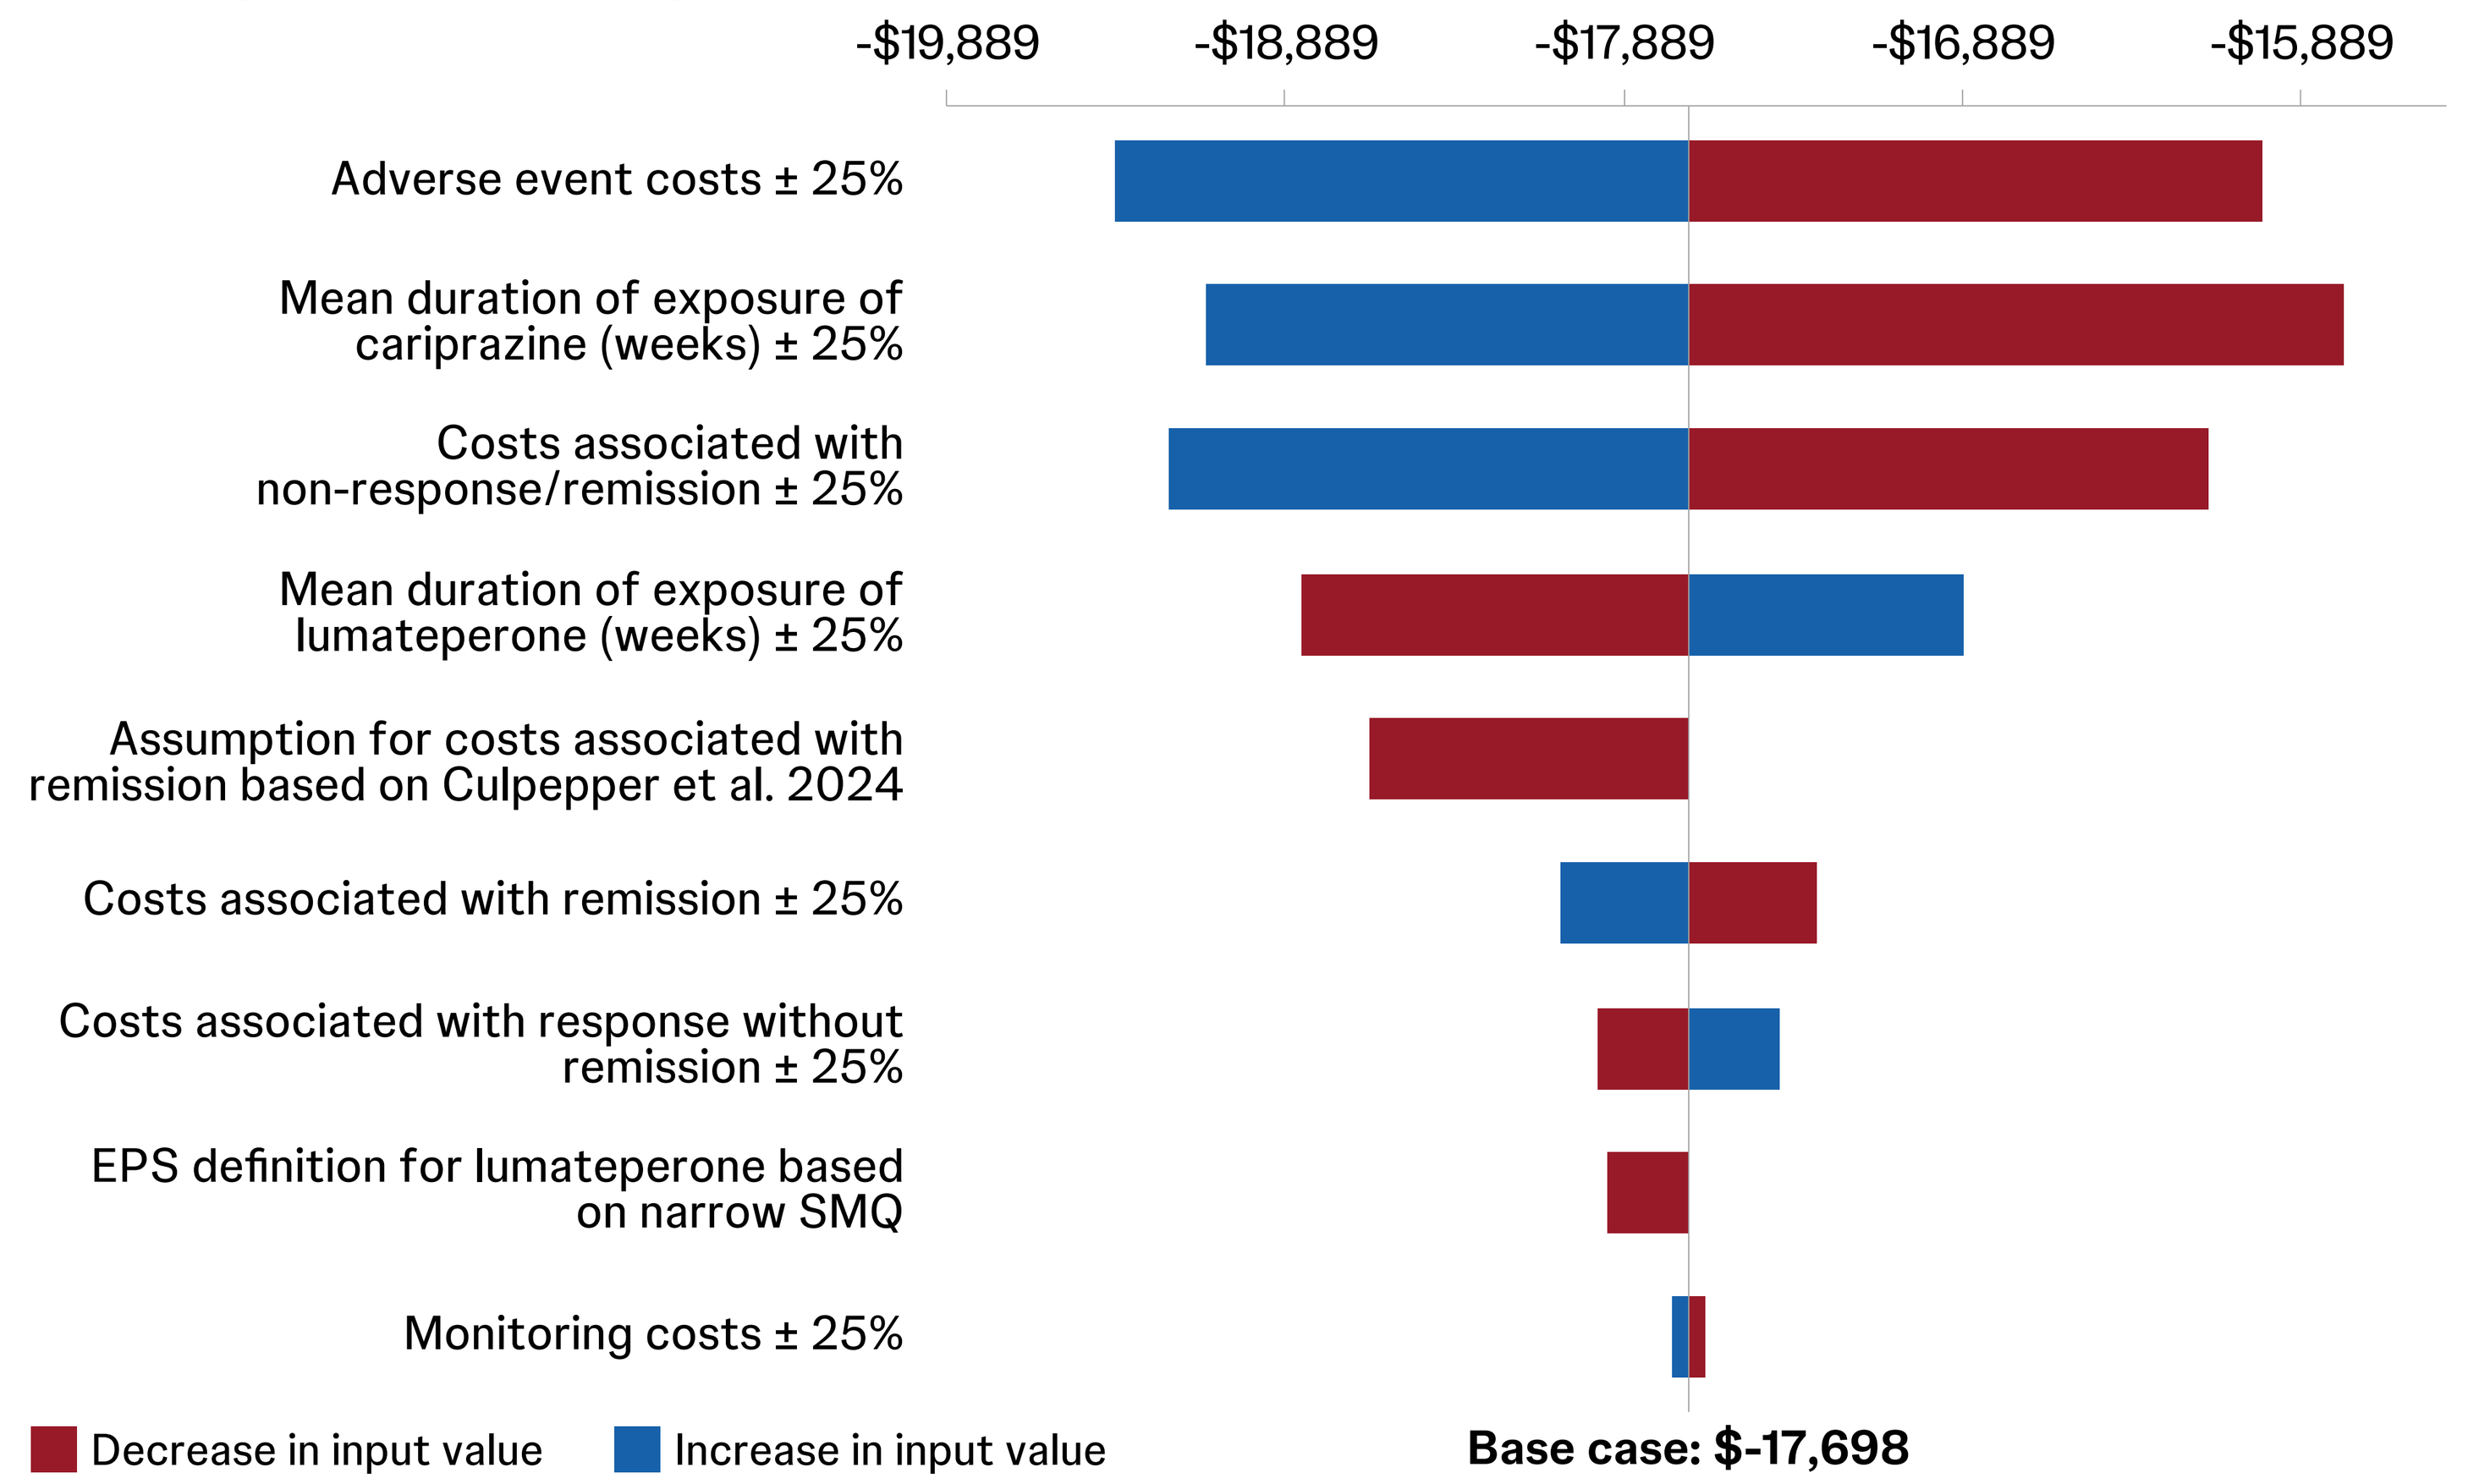
-$19,889
-$18,889
-$17,889
-$16,889
-$15,889
Adverse event costs ± 25%
Mean duration of exposure of cariprazine (weeks) ± 25%
Costs associated with non-response/remission ± 25%
Mean duration of exposure of lumateperone (weeks) ± 25%
Assumption for costs associated with remission based on Culpepper et al. 2024
Costs associated with remission ± 25%
Costs associated with response without remission ± 25%
EPS definition for lumateperone based on narrow SMQ
Monitoring costs ± 25%
Decrease in input value
Increase in input value
Base case: $-17,698

Abbreviations: EPS, extrapyramidal symptoms; SMQ, Standardised MedDRA Query.

Conclusions

- In this economic model, lumateperone demonstrated a lower cost per remitter than cariprazine, driven by lower total costs, higher remission rates, and lower modeled AE costs. Compared to cariprazine, lumateperone also demonstrated a lower cost per responder. These results remained consistent in scenario and sensitivity analyses, including alternative approaches for cariprazine efficacy outcomes
- When compared to total cost per remitter estimates for a range of MDD treatments by Malone et al. 2007 ($26,131-$33,172, in 2026 USD)[15], the total cost per remitter for lumateperone remained lower
- The results of this study suggest that lumateperone demonstrates favorable economic value as adjunctive treatment for adult patients with MDD from a US commercial payer perspective

Limitations

- No head-to-head trial or indirect treatment comparison was available for lumateperone versus cariprazine over the extended follow-up captured by their open-label extensions; efficacy and safety inputs were based on a naïve comparison of outcomes reported in the respective OLE studies
 - To address uncertainty in cariprazine efficacy estimates, the model evaluated both OC and LOCF approaches, and results favored lumateperone under both approaches, with the OC approach being used in the base case as a conservative assumption
- Although additional assumptions were made for model inputs, several scenario and sensitivity analyses were conducted to test the impact of individual parameters and assumptions, which showed the model to be robust

Acknowledgements

- This study was funded by Johnson & Johnson Innovative Medicine, Raritan, NJ, USA.

Disclosures

- MN is an employee of Johnson & Johnson Innovative Medicine.
- EW, MX, JE, HC, and ZYZ are employees of Analysis Group Inc., which received consulting fees from Johnson & Johnson Innovative Medicine for this study.

Mood Disorders

Scan the QR code

The QR code is intended to provide scientific information for individual reference, and the information should not be altered or reproduced in any way

Presented at: Psych Congress 2026, September 15 – 19, 2026.
New Orleans, Louisiana | USA

References

1. Greenberg PE, et al. *Pharmacoeconomics* 2021; 39(6): 653–65.
2. Rush AJ, et al. *Am J Psychiatry* 2006; 163(11): 1905–17.
3. Culpepper L, et al. *Adv Ther* 2024; 41(5): 1860–84.
4. Kim HK, et al. *J Affect Disord* 2024; 361: 651–8.
5. Durgam S, et al. *Eur Neuropsychopharmacol* 2026; 108: 112786.
6. Vieta E, et al. *Int Clin Psychopharmacol* 2019; 34(2): 76–83.
7. Merative Micromedex. RED BOOK. micromedexsolutions.com. Accessed May 2026.
8. CMS. Physician Fee Schedule 2026. cms.gov. Accessed May 2026.
9. CMS. Clinical Laboratory Fee Schedule, CY 2026 Q2. cms.gov. Accessed May 2026.
10. Dodd S, et al. *World J Biol Psychiatry* 2018; 19(5): 330–48.
11. CAPLYTA (lumateperone). Prescribing information. Intra-Cellular Therapies; April 2026.
12. VRAYLAR (cariprazine). Prescribing information. AbbVie; December 2025.
13. Kadakia A, et al. *J Med Econ* 2022; 25(1): 87–98.
14. Tkacz J, et al. *PharmacoEconomics Open* 2026; 10(3): 567–75.
15. Malone D. *J Manag Care Pharm* 2007; 13(6 Suppl): S8–18.

This material is distributed for scientific purposes by Johnson & Johnson Innovative Medicine and is not for promotional use.