112

Lumateperone 42 mg in Patients With Major Depressive Disorder: Analysis of Remission in Short- and Long-Term Trials

Michael E. Thase, MD[1]; Suresh Durgam, MD[2]; Willie R. Earley, MD[3]; Changzheng Chen, PhD[3]; Tobie Escher, PhD[3]

[1]Perelman School of Medicine, University of Pennsylvania, Philadelphia, PA, USA; [2]Former employee of Intra-Cellular Therapies, a Johnson & Johnson company, Bedminster, NJ, USA; [3]Intra-Cellular Therapies, a Johnson & Johnson company, Bedminster, NJ, USA

This material is distributed for scientific purposes by Johnson & Johnson Innovative Medicine and is not for promotional use.

Background

- Major depressive disorder (MDD) is a prevalent and complex psychiatric condition, affecting ≈185 million individuals worldwide[1]
- In patients with MDD, remission rates are low (≈25%) following first-line treatment and decline further with subsequent treatments[2]
- Failure to achieve remission is associated with reduced quality of life and increased relapse rates,[3] underscoring the importance of remission as a key goal of MDD treatment
- Lumateperone is an atypical antipsychotic indicated for the treatment of MDD in adults as adjunct to antidepressant therapy (ADT); the treatment of schizophrenia in adults; and the treatment of depressive episodes associated with bipolar I or II disorder (bipolar depression) in adults, as monotherapy and as adjunctive therapy with lithium or valproate[4]
 - Lumateperone is a simultaneous modulator of serotonin, dopamine, and glutamate neurotransmission[5]
 - Specifically, lumateperone is a potent serotonin 5-HT_{2A} receptor antagonist, a dopamine D_2 receptor presynaptic partial agonist and postsynaptic antagonist, a D_1 receptor-dependent indirect modulator of glutamatergic AMPA and NMDA currents, and a serotonin reuptake inhibitor[5]
 - This novel mechanism of action, characterized by its multimodal effects, may confer robust efficacy with improved tolerability compared with current treatment options
- The efficacy and safety of lumateperone 42 mg adjunctive to ADT was demonstrated in two Phase 3, randomized, double-blind, placebo-controlled studies (Study 501 [NCT04985942]; Study 502 [NCT05061706]) and an open-label extension study (OLE; Study 503 [NCT05061719]) in patients with MDD with inadequate ADT response[6-8]
 - The primary and key secondary endpoints were met in both short-term studies, with lumateperone 42 mg + ADT significantly improving Montgomery-Åsberg Depression Rating Scale (MADRS) Total score and Clinical Global Impression-Severity (CGI-S) score, respectively, vs placebo + ADT at Day 43; efficacy was maintained throughout the 26-week OLE
 - Lumateperone 42 mg + ADT was generally well tolerated in both short- and long-term studies
- This analysis of Studies 501, 502, and 503 evaluated MADRS Total score remission rates in patients with MDD with inadequate ADT response

Methods

- Efficacy data were pooled for the lumateperone 42 mg + ADT group and the placebo + ADT group from the short-term Studies 501/502; data were presented for the long-term Study 503
- Study 501 and Study 502 enrolled adults (18-65 years old) who met *Diagnostic and Statistical Manual of Mental Disorders, Fifth Edition* (DSM-5) criteria for MDD with inadequate response to 1-2 ADTs in the current depressive episode (defined as <50% improvement according to the Antidepressant Treatment Response Questionnaire)
 - Patients were experiencing a major depressive episode (MADRS Total score ≥24, CGI-S score ≥4) and had Quick Inventory of Depressive Symptomatology-Self Report-16 item score ≥14 at screening and baseline
 - Patients were randomized 1:1 to 6-week, double-blind placebo + ADT or lumateperone 42 mg + ADT
- Patients who safely completed double-blind treatment could enroll in Study 503 to receive 26-week, open-label, oral, once-daily lumateperone 42 mg + ADT
 - Screening for the OLE occurred on the last visit of the lead-in study (lead-in study Day 43, OLE Day 1)
- In pooled Studies 501/502 and Study 503, efficacy was evaluated in the overall populations and in patient subgroups (age, type of ADT, and baseline MADRS severity), using logistic regression in the placebo-controlled studies and descriptive statistics in the OLE
- MADRS remission (MADRS Total score ≤10), complete remission (MADRS Total score ≤5), and sustained remission (defined at each visit as MADRS Total score ≤10 at the visit and MADRS Total score ≤10 maintained at all following visits) were assessed

Results

Patient Population

- The modified intent-to-treat (mITT) population comprised 950 patients (lumateperone + ADT, n=471; placebo + ADT, n=479) in the pooled Studies 501/502
- In Study 503, 809 patients were enrolled and treated with lumateperone + ADT
- For Studies 501/502 and Study 503, the majority of patients were female (67.7% and 68.9%, respectively) and White (86.0% and 88.2%, respectively)

Efficacy

- In pooled Studies 501/502, MADRS Total score remission rates were significantly greater (P<.0001) with lumateperone + ADT (25.5%) vs placebo + ADT (13.6%) at Day 43
- Significantly greater MADRS remission rates were observed in patient subgroups with lumateperone + ADT vs placebo + ADT in the pooled population at Day 43 (**Figure 1**)

Figure 1. MADRS Total Score Remission[a] Rates at Day 43 in Patient Subgroups: Pooled Studies 501/502

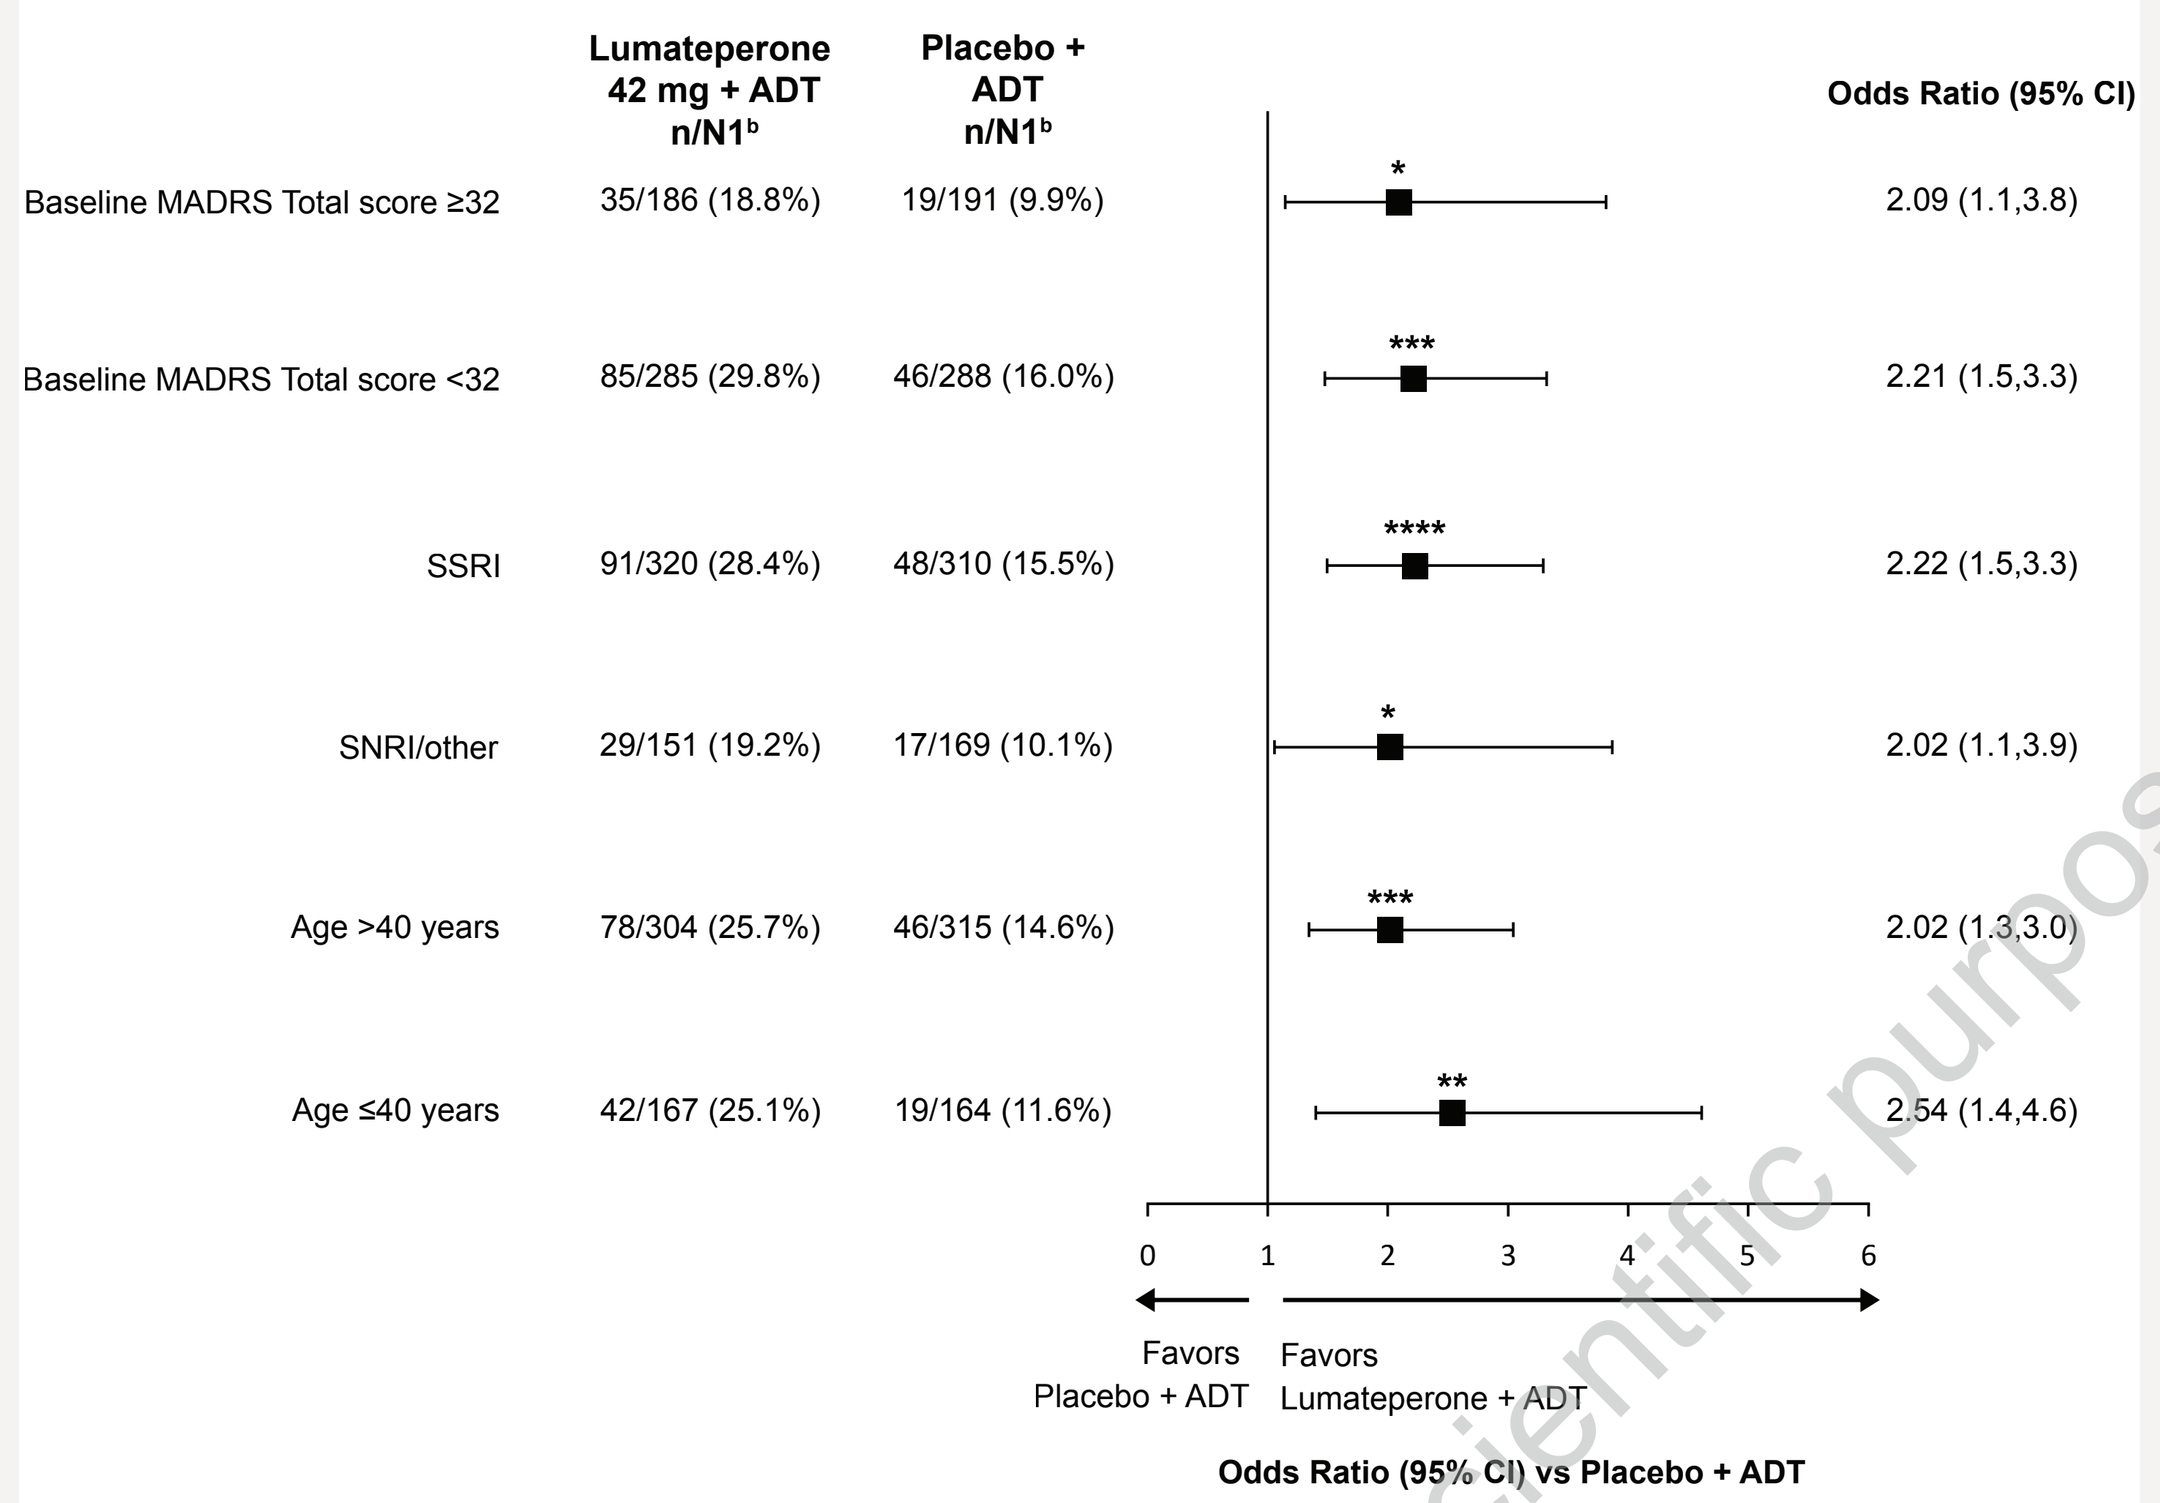
Lumateperone 42 mg + ADT n/N1[b]
Placebo + ADT n/N1[b]
Odds Ratio (95% CI)
Baseline MADRS Total score ≥32 | 35/186 (18.8%) | 19/191 (9.9%) | * | 2.09 (1.1,3.8)
Baseline MADRS Total score <32 | 85/285 (29.8%) | 46/288 (16.0%) | *** | 2.21 (1.5,3.3)
SSRI | 91/320 (28.4%) | 48/310 (15.5%) | **** | 2.22 (1.5,3.3)
SNRI/other | 29/151 (19.2%) | 17/169 (10.1%) | * | 2.02 (1.1,3.9)
Age >40 years | 78/304 (25.7%) | 46/315 (14.6%) | *** | 2.02 (1.3,3.0)
Age ≤40 years | 42/167 (25.1%) | 19/164 (11.6%) | ** | 2.54 (1.4,4.6)
0 1 2 3 4 5 6
Favors Placebo + ADT
Favors Lumateperone + ADT
Odds Ratio (95% CI) vs Placebo + ADT

*P<.05 **P<.01 ***P<.001 ****P<.0001. Logistic regression model in the mITT population.
[a]Remission was defined as MADRS Total score ≤10 at the visit. [b]N1=Number of patients in each subgroup.
ADT, antidepressant therapy; MADRS, Montgomery-Åsberg Depression Rating Scale; mITT, modified intent-to-treat; SNRI, serotonin-norepinephrine reuptake inhibitor; SSRI, selective serotonin reuptake inhibitor.

- Remission occurred in 529 (65.4%) patients at end of open-label treatment in Study 503
- In each of the subgroups analyzed in Study 503, MADRS Total score remission occurred in over 55% of patients receiving lumateperone + ADT at end of treatment (**Figure 2**)
 - Remission rates were similar regardless of age, ADT treatment, or baseline disease severity

Figure 2. MADRS Total Score Remission[a] Rates at EOT in Patient Subgroups: Study 503

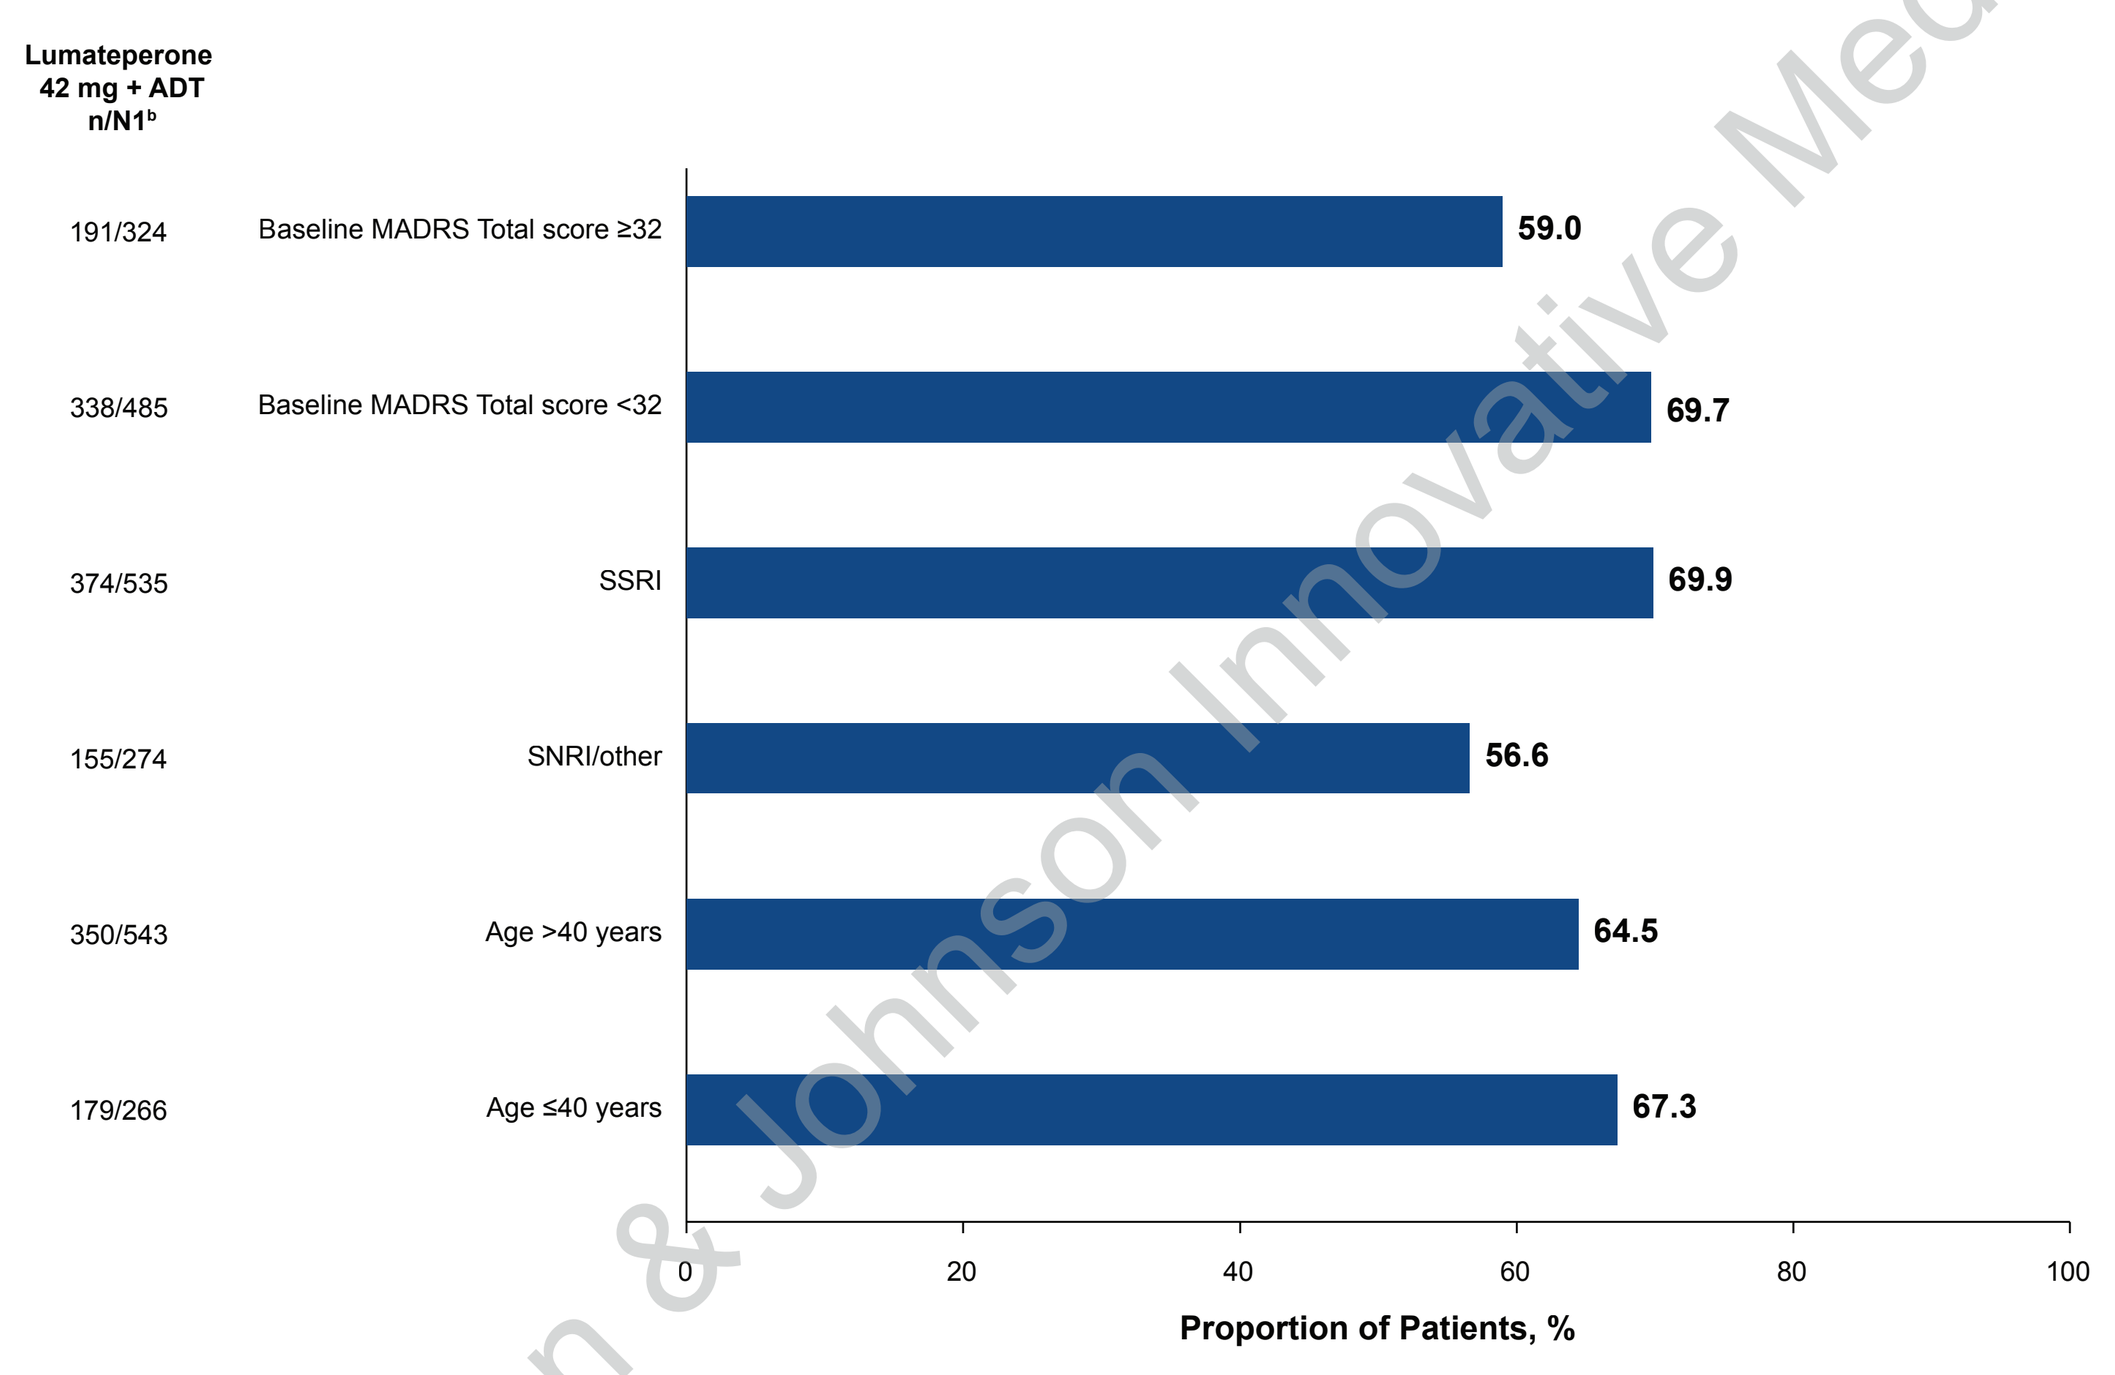
Lumateperone 42 mg + ADT n/N1[b]
191/324 Baseline MADRS Total score ≥32 59.0
338/485 Baseline MADRS Total score <32 69.7
374/535 SSRI 69.9
155/274 SNRI/other 56.6
350/543 Age >40 years 64.5
179/266 Age ≤40 years 67.3
0 20 40 60 80 100
Proportion of Patients, %

[a]Remission was defined as MADRS Total score ≤10 at the visit. [b]N1=Number of patients in each subgroup in the safety population.
ADT, antidepressant therapy; EOT, end of treatment; MADRS, Montgomery-Åsberg Depression Rating Scale; SNRI, serotonin-norepinephrine reuptake inhibitor; SSRI, selective serotonin reuptake inhibitor.

- Complete remission was achieved by 50 patients (10.6%) with lumateperone + ADT and 27 patients (5.6%) with placebo + ADT (P<.01) in the pooled mITT population of Studies 501/502 (**Figure 3A**)
- In Study 503, complete remission was achieved by 357 patients (44.1%) (**Figure 3B**)

Figure 3. MADRS Total Score Complete Remission[a]

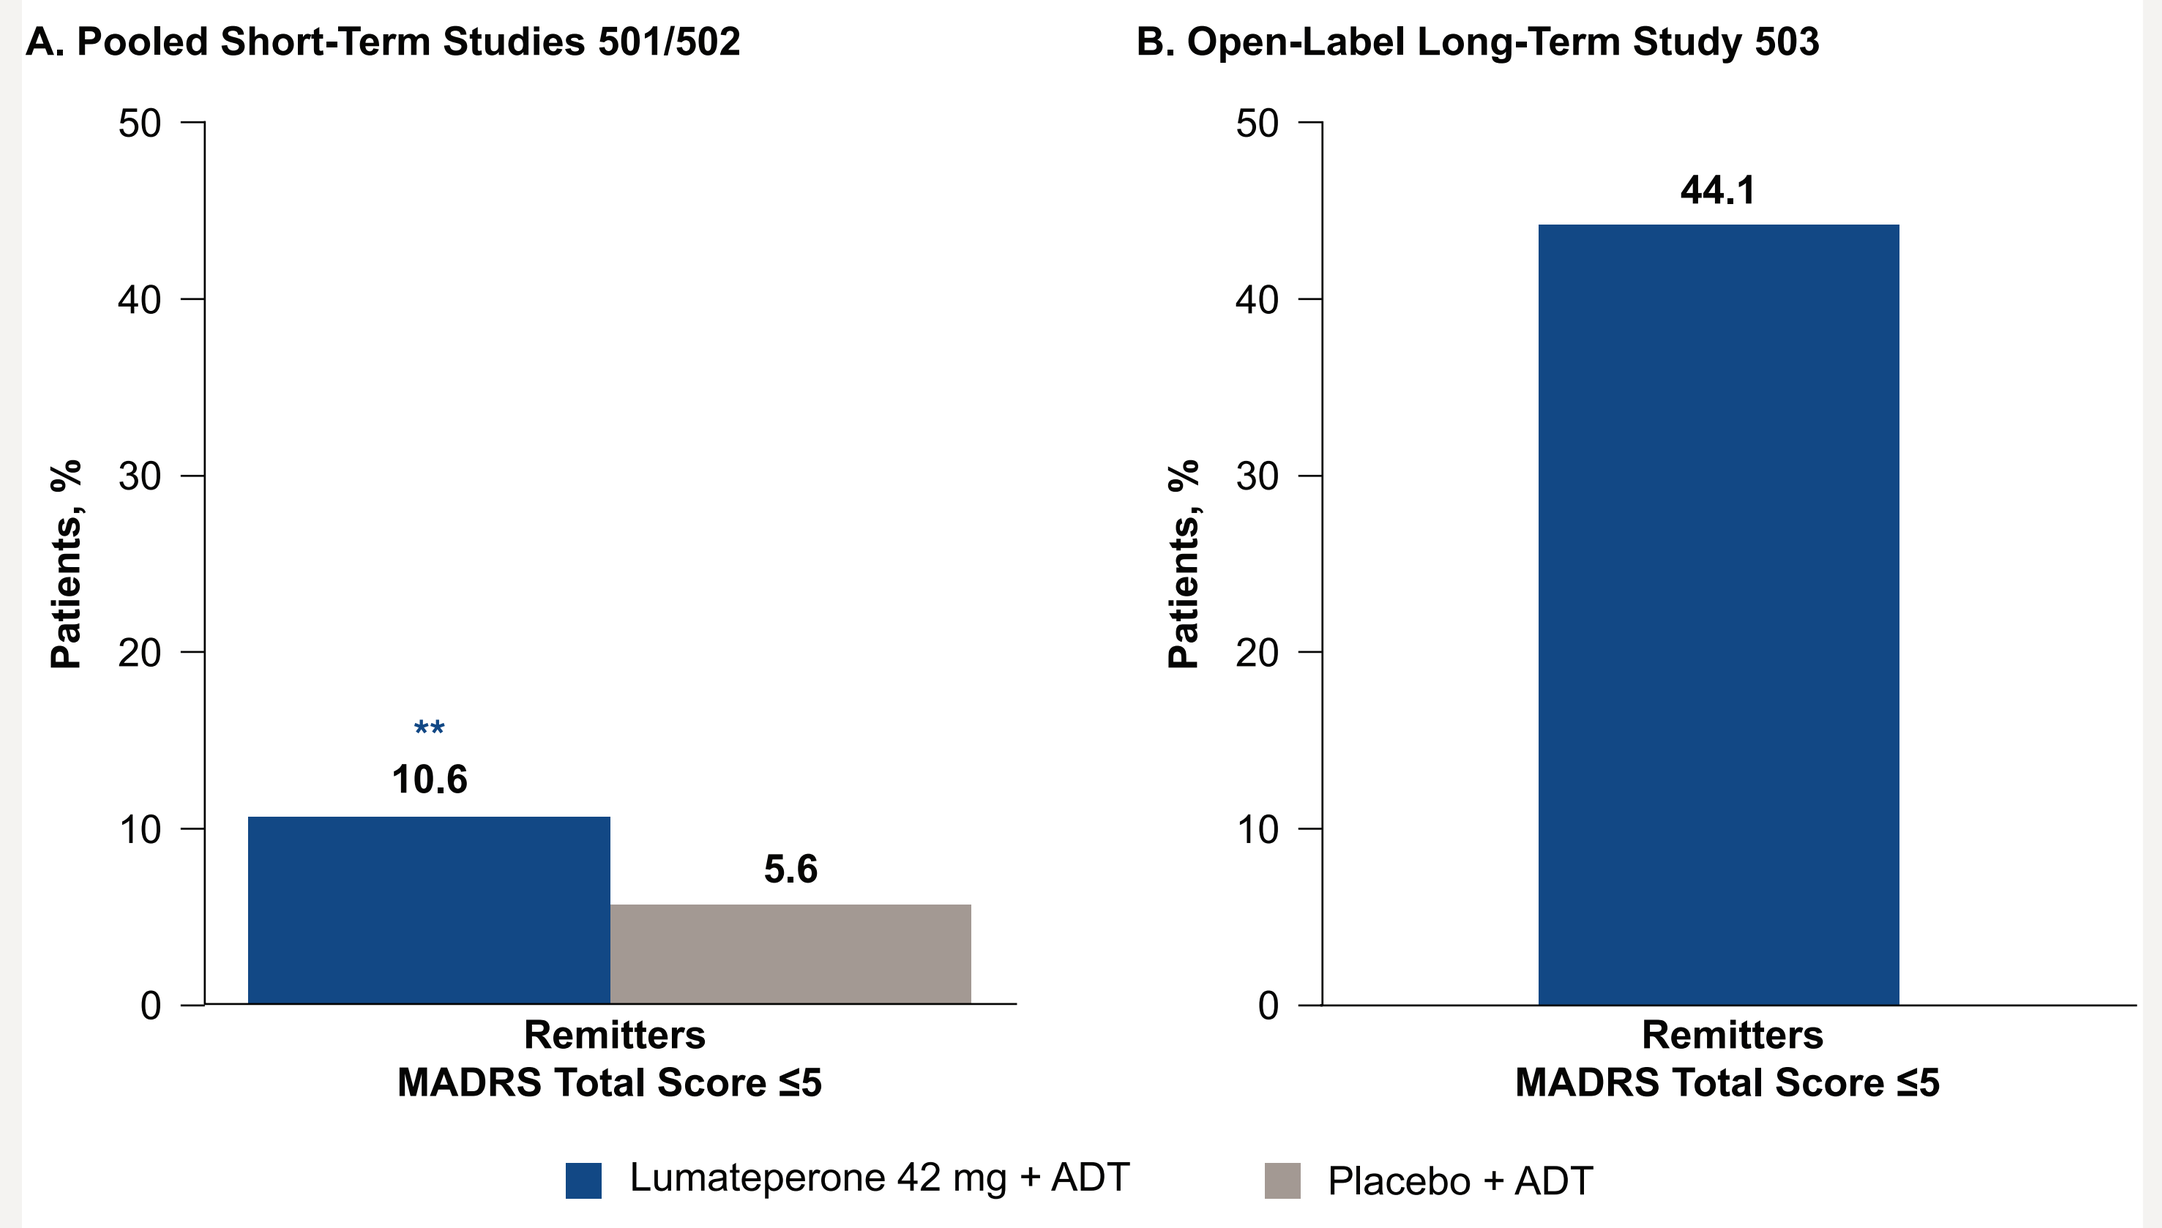
A. Pooled Short-Term Studies 501/502
B. Open-Label Long-Term Study 503
Patients, %
**
10.6
5.6
44.1
Remitters MADRS Total Score ≤5
Lumateperone 42 mg + ADT
Placebo + ADT

**P<.01. Logistic regression model in Studies 501/502.
[a]Complete remission was defined as MADRS Total score ≤5 at Day 43 in Studies 501/502 and end of open-label treatment in Study 503.
ADT, antidepressant therapy; MADRS, Montgomery-Åsberg Depression Rating Scale.

- Sustained remission rates were significantly greater with lumateperone + ADT vs placebo + ADT beginning at Day 22 in the pooled mITT population of Studies 501/502 (**Figure 4A**)
- In Study 503, sustained remission rates ranged from 13.2% at Week 1 to 60.4% at Week 26 (**Figure 4B**)

Figure 4. MADRS Total Score Sustained Remission[a] Rates

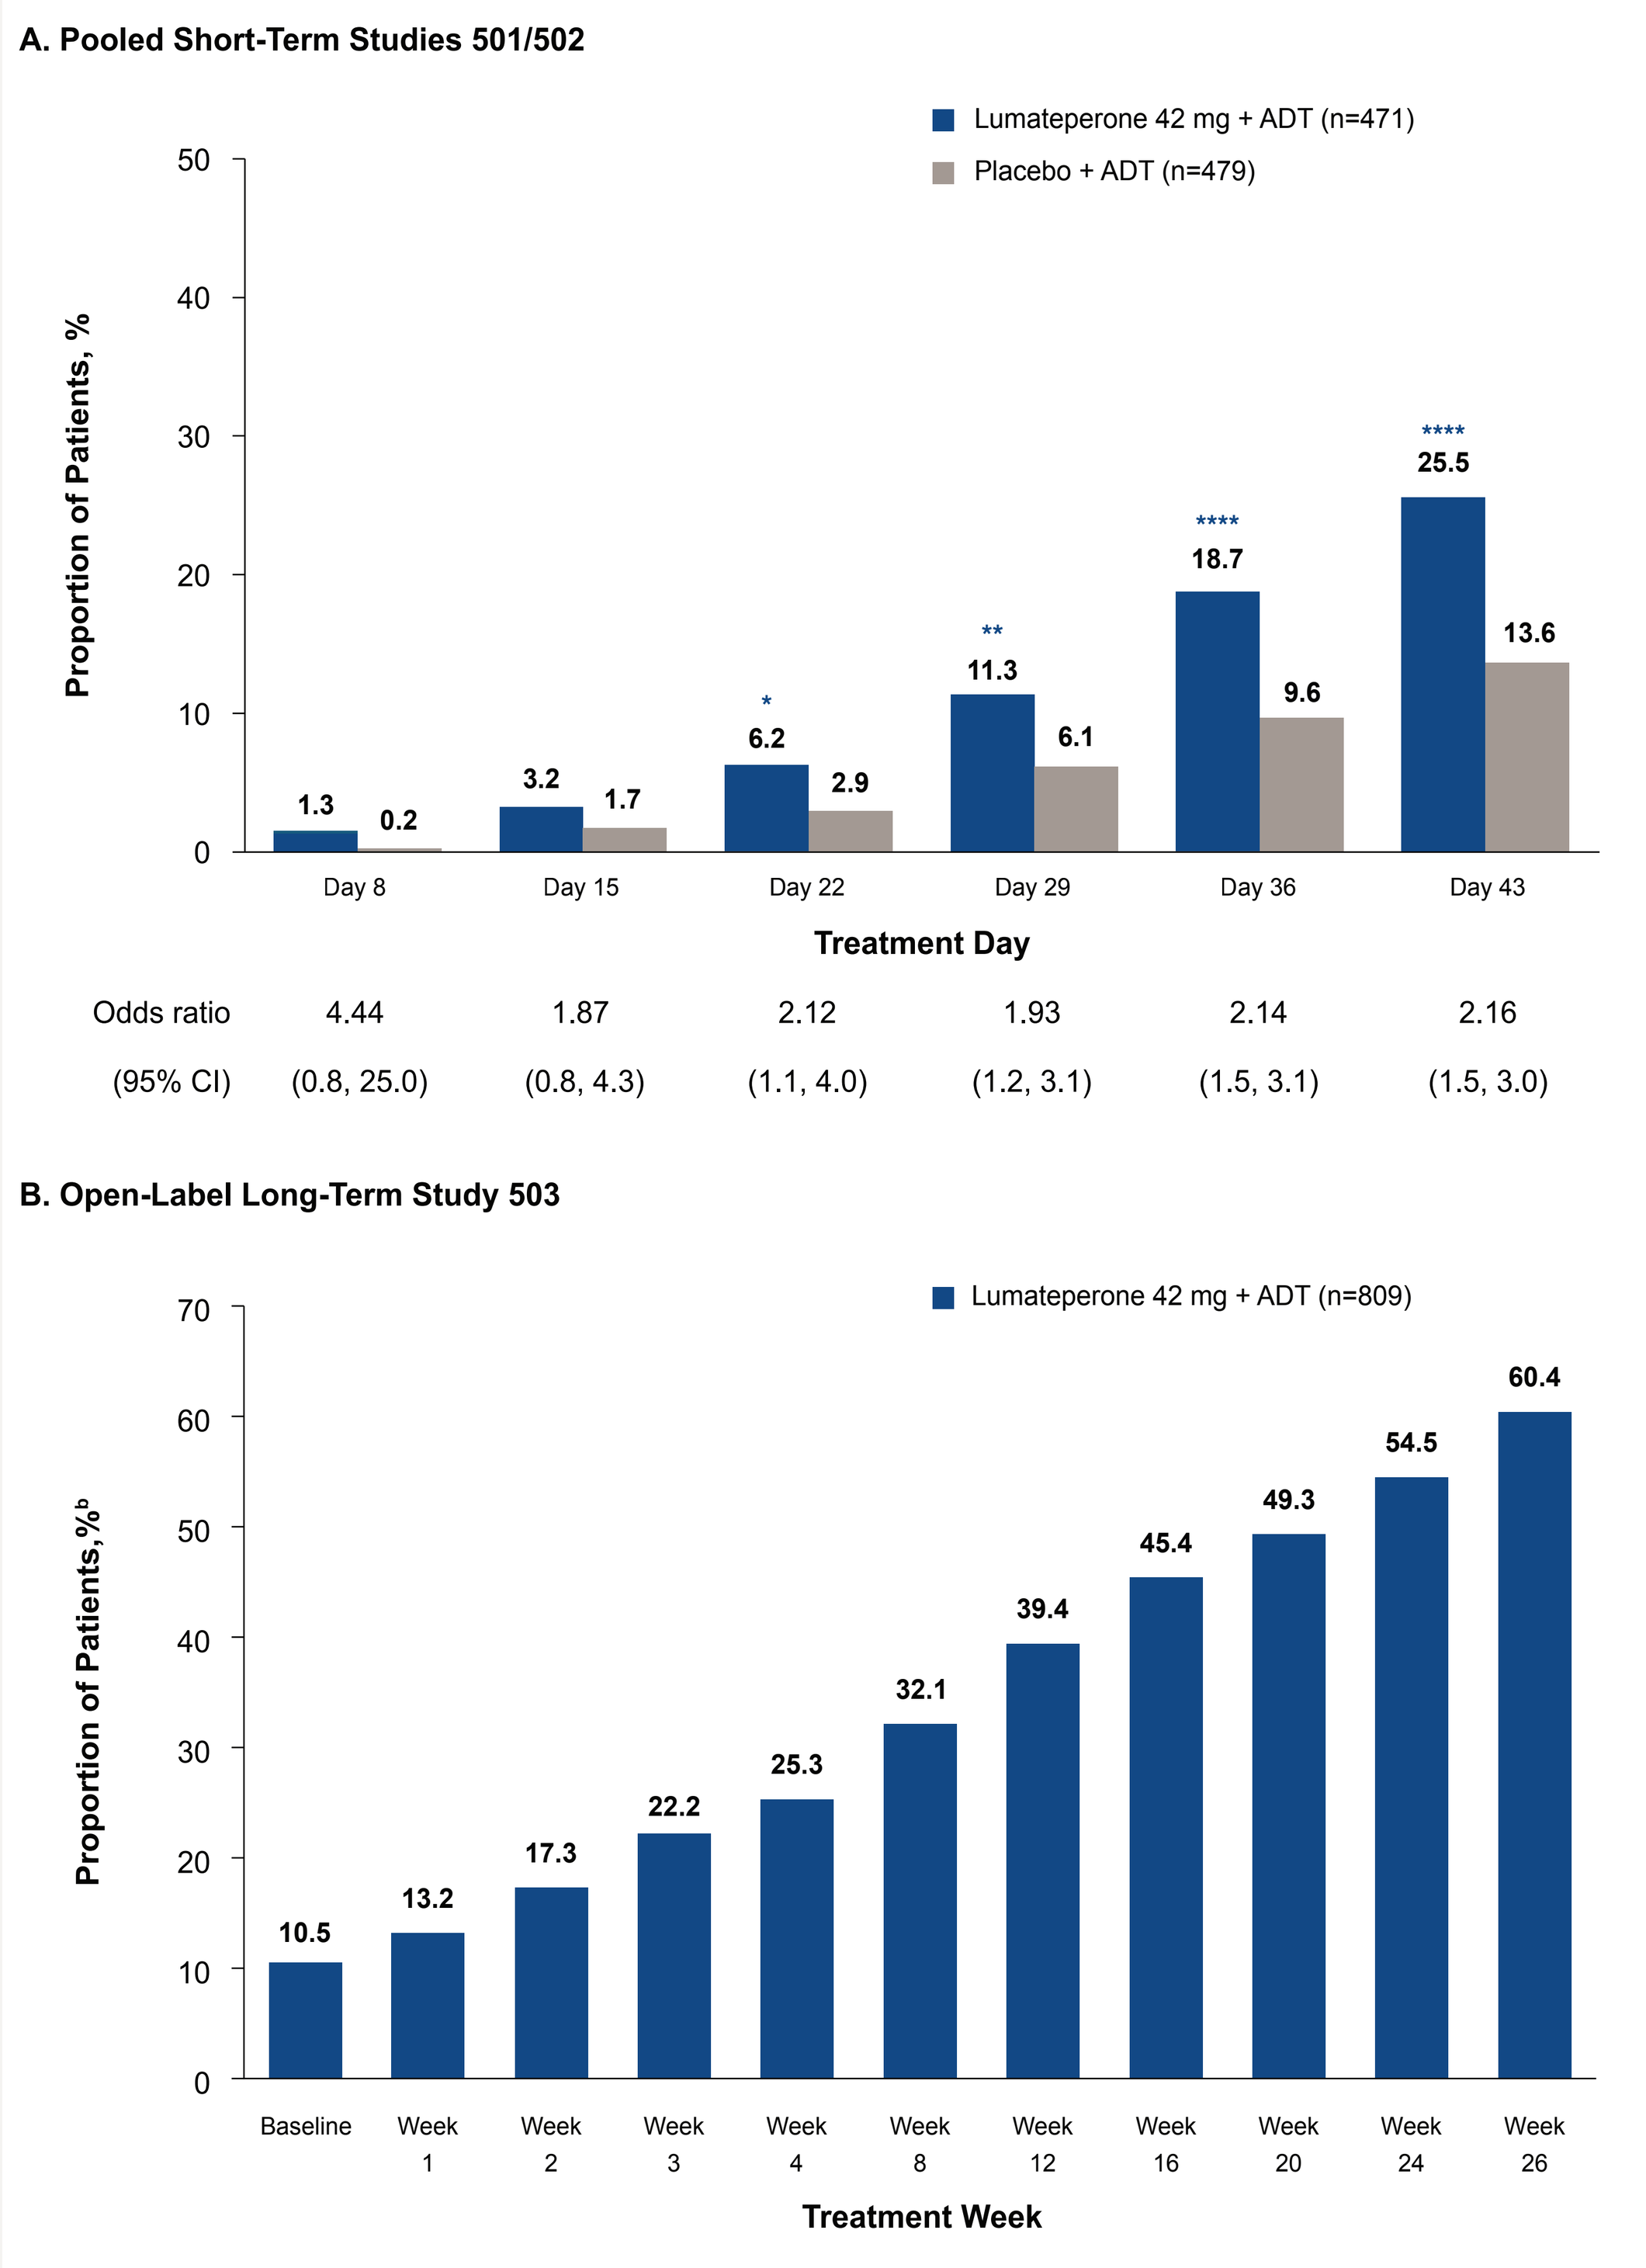
A. Pooled Short-Term Studies 501/502
Lumateperone 42 mg + ADT (n=471)
Placebo + ADT (n=479)
Proportion of Patients, %
Day 8: 1.3 / 0.2
Day 15: 3.2 / 1.7
Day 22: * 6.2 / 2.9
Day 29: ** 11.3 / 6.1
Day 36: **** 18.7 / 9.6
Day 43: **** 25.5 / 13.6
Treatment Day
Odds ratio 4.44 1.87 2.12 1.93 2.14 2.16
(95% CI) (0.8, 25.0) (0.8, 4.3) (1.1, 4.0) (1.2, 3.1) (1.5, 3.1) (1.5, 3.0)
B. Open-Label Long-Term Study 503
Lumateperone 42 mg + ADT (n=809)
Proportion of Patients, %[b]
Baseline 10.5
Week 1 13.2
Week 2 17.3
Week 3 22.2
Week 4 25.3
Week 8 32.1
Week 12 39.4
Week 16 45.4
Week 20 49.3
Week 24 54.5
Week 26 60.4
Treatment Week

*P<.05 **P<.01 ****P<.0001. Logistic regression model in the mITT population of pooled Studies 501/502.
[a]Sustained remission was defined at each visit as MADRS Total score ≤10 at the visit and MADRS Total score ≤10 maintained at all following visits.
[b]Percentage based on the safety population consisting of patients who safely completed the double-blind lead-in studies 501 or 502.
ADT, antidepressant therapy; MADRS, Montgomery-Åsberg Depression Rating Scale; mITT, modified intent-to-treat.

Conclusions

- Lumateperone 42 mg + ADT demonstrated significantly greater MADRS Total score remission rates over placebo + ADT in pooled short-term studies in patients with MDD with inadequate ADT response
- Efficacy was maintained with long-term lumateperone 42 mg + ADT treatment, with ≈2 of every 3 patients achieving remission with 6-month treatment
- These results indicate lumateperone as a promising adjunctive treatment option for patients with MDD with inadequate ADT response

Acknowledgments

The authors thank all study investigators, research staff, and patients for their participation. Medical writing support was provided by Thato Gweja, PhD, of Nucleus Global, an Inizio company, funded by Intra-Cellular Therapies, a Johnson & Johnson company.

Disclosures

ME Thase has served as an advisor or a consultant for Autobahn Therapeutics; Axsome Therapeutics, Inc.; Clexio Biosciences; Gerson Lehman; GH Therapeutics; H. Lundbeck, A/S; Janssen Pharmaceuticals, Inc.; Johnson & Johnson; Luye Pharma Group, Ltd.; Merck & Company, Inc.; Object Pharma; Otsuka Pharmaceutical Company, Ltd.; Pfizer, Inc.; Sage Pharmaceuticals; Seelos Pharmaceuticals; Takeda Pharmaceutical Company, Ltd.; has received grants from Acadia Inc.; Alkermes; Axsome Therapeutics Inc.; Intra-Cellular, Inc.; Janssen Pharmaceuticals, Inc.; Myriad; National Institute of Mental Health; Otsuka Pharmaceutical Company, Ltd.; Patient-Centered Outcomes Research Institute (PCORI); Takeda Pharmaceutical Company, Ltd.; and has received royalties from the American Psychiatric Foundation; Guilford Publications; Herald House; Wolters Kluwer; W.W. Norton & Company, Inc., and spouse's employment with Open Health, which does business with most major pharmaceutical companies.

S Durgam is a former employee of Intra-Cellular Therapies, a Johnson & Johnson company.

WR Earley, C Chen, and T Escher are full-time employees of Intra-Cellular Therapies, a Johnson & Johnson company.

Funding

This study was funded by Intra-Cellular Therapies, a Johnson & Johnson company.

Previously presented at the 64th Annual Meeting of the American College of Neuropsychopharmacology (ACNP), January 12-15, 2026, Nassau, Bahamas.

Neuropsychiatry

Scan the QR code

The QR code is intended to provide scientific information for individual reference, and the information should not be altered or reproduced in any way.

Email questions to
Willie R. Earley at wearley@its.jnj.com

Presented at Psych Congress 2026,
September 15-19, 2026, New Orleans, LA, USA

References:
1. Marx W, et al. *Nat Rev Dis Primers.* 2023;9(1):44; 2. Pigott HE, et al. *BMJ Open.* 2023;13:e063095; 3. IsHak WW, et al. *Acta Psychiatr Scand.* 2015;131:51-60; 4. Caplyta (lumateperone). Prescribing information. Intra-Cellular Therapies, Inc; 2026; 5. Titulaer J, et al. *Eur Neuropsychopharmacol.* 2022;62:22-35; 6. Durgam S, et al. *J Clin Psychiatry.* 2025;86(4):25m15848; 7. Durgam S, et al. *Am J Psychiatry.* 2025;182(12):1072-1082; 8. Durgam S, et al. *Eur Neuropsychopharmacol.* 2026. 108:112786.