82

Preservation of Cardiometabolic Health During Adjunctive Lumateperone Treatment in Major Depressive Disorder (MDD): A Post Hoc Analysis

Andrew Krystal, MD[1]; C. Neill Epperson, MD[2]; Ibrahim Turkoz, PhD[3]; Brian Curcio, MS[4]; Kristin R. Dorsey, Pharm, D[3]; Zhiheng Zhang, MS[3]; Christoph U. Correll, MD[5-9]

[1]Departments of Psychiatry and Neurology, University of California San Francisco School of Medicine, San Francisco, CA, USA; [2]Department of Psychiatry, University of Colorado School of Medicine, Anschutz Medical Campus, Aurora, CO, USA; [3]Johnson & Johnson, Titusville, NJ, USA; [4]ICON plc. Philadelphia, PA, USA; [5]Department of Psychiatry, Zucker Hillside Hospital, Northwell Health, Glen Oaks, NY, USA; [6]Department of Psychiatry and Molecular Medicine, Zucker School of Medicine at Hofstra/Northwell, Hempstead, NY, USA; [7]Center for Psychiatric Neuroscience, Feinstein Institute for Medical Research, Manhasset, NY, USA; [8]Department of Child and Adolescent Psychiatry, Charité Universitätsmedizin, Berlin, Germany; [9]German Center for Mental Health (DZPG), Partner Site Berlin, Berlin, Germany

This material is distributed for scientific purposes by Johnson & Johnson Innovative Medicine and is not for promotional use.

Background

- Major depressive disorder (MDD) is a complex and highly burdensome mental illness associated with functional impairment, reduced quality of life, and comorbidities including cardiovascular disease, diabetes, and metabolic syndrome (MetSy)[1,2]
- MetSy is defined as meeting 3 of the following 5 criteria[3]:
 - Waist circumference >40 in (men) or >35 in (women)
 - Triglycerides ≥150 mg/dL
 - High density lipoprotein (HDL) cholesterol <40 mg/dL (men) or <50 mg/dL (women)
 - Systolic blood pressure (BP) ≥130 mm Hg or diastolic BP ≥85 mm Hg
 - Fasting glucose ≥100 mg/dL
- Patients with MetSy have an elevated risk of developing type 2 diabetes and increased mortality due to cardiovascular disease[4]
- Lumateperone is FDA approved for the treatment of MDD in adults as adjunct to antidepressant therapy (ADT); treatment of schizophrenia in adults; and treatment of depressive episodes associated with bipolar I or II disorder (bipolar depression) in adults, as monotherapy and as adjunctive therapy with lithium or valproate[5]
 - Lumateperone modulates serotonergic, dopaminergic, and indirectly glutamatergic neurotransmission[5,6]
 - Lumateperone acts as a potent antagonist at 5-HT$_{2A}$ receptors, a moderate antagonist at postsynaptic D$_2$ receptors and a presynaptic partial agonist at D$_2$ receptors, an indirect modulator of glutamate neurotransmission through enhancement of NMDA receptor function and upregulation of AMPA receptor activity via the mTOR pathway, and a SERT inhibitor[7]
 - This novel mechanism of action with multimodal effects may confer robust efficacy with improved safety and tolerability compared with current treatment options
- In 2 Phase 3, randomized, double-blind, placebo-controlled studies (Study 501, NCT04985942; Study 502, NCT05061706), adjunctive lumateperone 42 mg provided superior efficacy vs adjunctive placebo, with meaningful and significant improvements in depressive symptoms (Montgomery-Åsberg Depression Rating Scale [MADRS] Total score) and disease severity (Clinical Global Impression–Severity [CGI-S] score)[8,9]
- In a long-term open-label extension study (OLE; Study 503 [NCT05061719]) in patients with MDD improvements in depressive symptoms and disease severity with adjunctive lumateperone 42 mg treatment continued from the double-blind study baseline to the end of the 6-month open-label treatment period[10]
- Lumateperone 42 mg + ADT was generally well tolerated in both individual short- and long-term MDD studies[8-10]
 - In the short- and long-term Phase 3 trials, changes in weight and cardiometabolic parameters were not clinically meaningful in the adjunctive lumateperone group and were generally similar to those in the adjunctive placebo group in the short-term trials
- Many ADTs are associated with treatment-related effects such as weight gain, hyperlipidemia, and hyperglycemia, increasing MetSy risk and highlighting the need for treatments with a more favorable metabolic profile[11,12]
- This analysis evaluated the effect of short-term (pooled analysis of Studies 501 and 502) and long-term (OLE Study 503) treatment with adjunctive lumateperone 42 mg on cardiometabolic parameters, including MetSy and 10-year cardiovascular risk estimates, in patients with MDD

Methods

- Studies 501 and 502 enrolled adults (18-65 years old) who met *Diagnostic and Statistical Manual of Mental Disorders, Fifth Edition* (DSM-5) criteria for MDD with inadequate response to 1-2 ADTs in the current depressive episode (defined as <50% improvement according to the Antidepressant Treatment Response Questionnaire)[8,9]
 - Patients were experiencing a major depressive episode (MADRS Total score ≥24, CGI-S score ≥4) and had Quick Inventory of Depressive Symptomatology-Self Report-16 item score ≥14 at screening and baseline
 - Patients were randomized 1:1 to 6-week, double-blind placebo + ADT or lumateperone 42 mg + ADT
- Patients who completed double-blind treatment could enroll in Study 503 to receive 26-week, open-label, oral, once-daily lumateperone 42 mg + ADT[10]
 - Screening for the OLE occurred on the last visit of the lead-in study (lead-in study Day 43, OLE Day 1)
- This post hoc analysis evaluated MetSy status at baseline and end of treatment (EOT) as well as the percentage of patients who converted MetSy status (Yes to No and No to Yes) from baseline to EOT for the pooled 501/502 studies and the OLE 503 study; the following were analyzed:
 - Overall population
 - Stratified by concomitant ADT (SSRI vs SNRI)
- Change in individual MetSy criteria from baseline to EOT was also analyzed for the pooled 501 and 502 and OLE studies
- Given that Hb$_{A1c}$ is a measure of average blood glucose over the past 2 to 3 months, Hb$_{A1c}$ status (≤7% vs >7%) from baseline to EOT was assessed for the OLE study
- Additionally, long-term cardiovascular impact was estimated at baseline and EOT using the Framingham general cardiovascular disease risk model published by D'Agostino et al[13]
 - Smoking status data were not available for all patients, therefore this analysis was conducted assuming that all patients were non-smokers and smokers

Results

Patient Population

- The pooled safety population from studies 501 and 502 comprised 964 patients (lumateperone 42 mg + ADT, n=483; placebo + ADT, n=481); the OLE population comprised 809 patients
- Baseline characteristics were similar between treatment arms (**Table 1**)
 - Approximately one-third of patients had MetSy at baseline in the pooled short-term studies

Table 1. Baseline Demographics and Clinical Characteristics (mITT Population)

	Pooled Short-Term Studies (Studies 501 and 502)[8,9]		Open-Label Long-Term Study[10]
	Lumateperone 42 mg + ADT (n=483)	Placebo + ADT (n=481)	Lumateperone 42 mg + ADT (n=809)
Age, mean (range), years	45.3 (18-65)	45.8 (18-65)	46.2 (18-66)
Female, n (%)	327 (67.7)	325 (67.6)	549 (67.9)
Race, %			
White	415 (85.9)	414 (86.1)	703 (86.9)
Asian	41 (8.5)	36 (7.5)	61 (7.5)
Black	26 (5.4)	24 (5.0)	37 (4.6)
Other	1 (0.2)	7 (1.5)	8 (1.0)
Weight, mean (SD), kg [N]	78.5 (16.7) [483]	79.0 (17.2) [481]	79.0 (16.9) [790]
BMI, mean (SD), kg/m² [N]	27.8 (5.0) [483]	27.8 (5.1) [481]	27.8 (5.0) [778]
≥30 (obese), n (%)	141 (29.2)	163 (33.9)	261 (32.3)
Waist circumference, mean (SD), cm [N]	92.5 (13.5) [482]	92.8 (14.0) [481]	93.1 (13.7) [808]
Men	96.1 (12.1) [156]	98.5 (12.2) [156]	97.5 (12.5) [260]
Women	90.7 (13.8) [326]	90.0 (14.0) [325]	90.9 (13.8) [548]
Triglycerides, mean (SD), mg/dL [N]	139.7 (89.1) [438]	133.5 (75.0) [466]	137.3 (81.7) [778]
HDL cholesterol, mean (SD), mg/dL [N]	56.2 (17.3) [438]	57.4 (16.5) [466]	56.7 (16.9) [778]
HDL cholesterol <40 mg/dL (men), n (%) [N]	47 (33.1) [142]	33 (30.0) [110]	67 (31.8) [211]
HDL cholesterol <50 mg/dL (women), n (%) [N]	95 (66.9) [142]	77 (70.0) [110]	144 (68.2) [211]
Supine systolic blood pressure, mean (SD), mmHg [N]	121.6 (10.5) [467]	122.6 (10.9) [479]	122.3 (10.5) [809]
Supine diastolic blood pressure, mean (SD), mmHg [N]	77.4 (7.8) [467]	77.8 (7.7) [479]	78.1 (7.5) [809]
Fasting glucose, mean (SD), mg/dL [N]	92.3 (14.7) [435]	93.7 (15.5) [463]	93.3 (14.8) [774]
MetSy at baseline, n (%)	158 (32.7)	162 (33.7)	267 (33.0)
Hb$_{A1c}$, median (range), % [N]	5.5 (4-7) [481]	5.5 (4-8) [480]	5.5 (4-8) [807]
ADT used during double-blind treatment, n (%)			
SSRI	328 (67.9)	312 (64.9)	535 (66.1)
SNRI	126 (26.1)	138 (26.7)	224 (27.7)
Other (bupropion)	29 (6.0)	31 (6.4)	50 (6.2)

ADT, antidepressant therapy; BMI, body-mass index; HDL, high-density lipoprotein; MetSy, metabolic syndrome; SD, standard deviation; SNRI, serotonin-norepinephrine reuptake inhibitor; SSRI, selective serotonin reuptake inhibitor.

Metabolic Syndrome: Overall Population

- In the pooled short-term studies, the incidence of patients with MetSy decreased from double-blind baseline to EOT with lumateperone 42 mg + ADT and increased from double-blind baseline to EOT with placebo + ADT (**Figure 1A**)
- The rate of MetSy decreased from OLE baseline to EOT (**Figure 1B**)

Figure 1. Metabolic Syndrome at Baseline and End of Treatment in Pooled Short-Term (A) and Long-Term (B) Studies: Overall Population

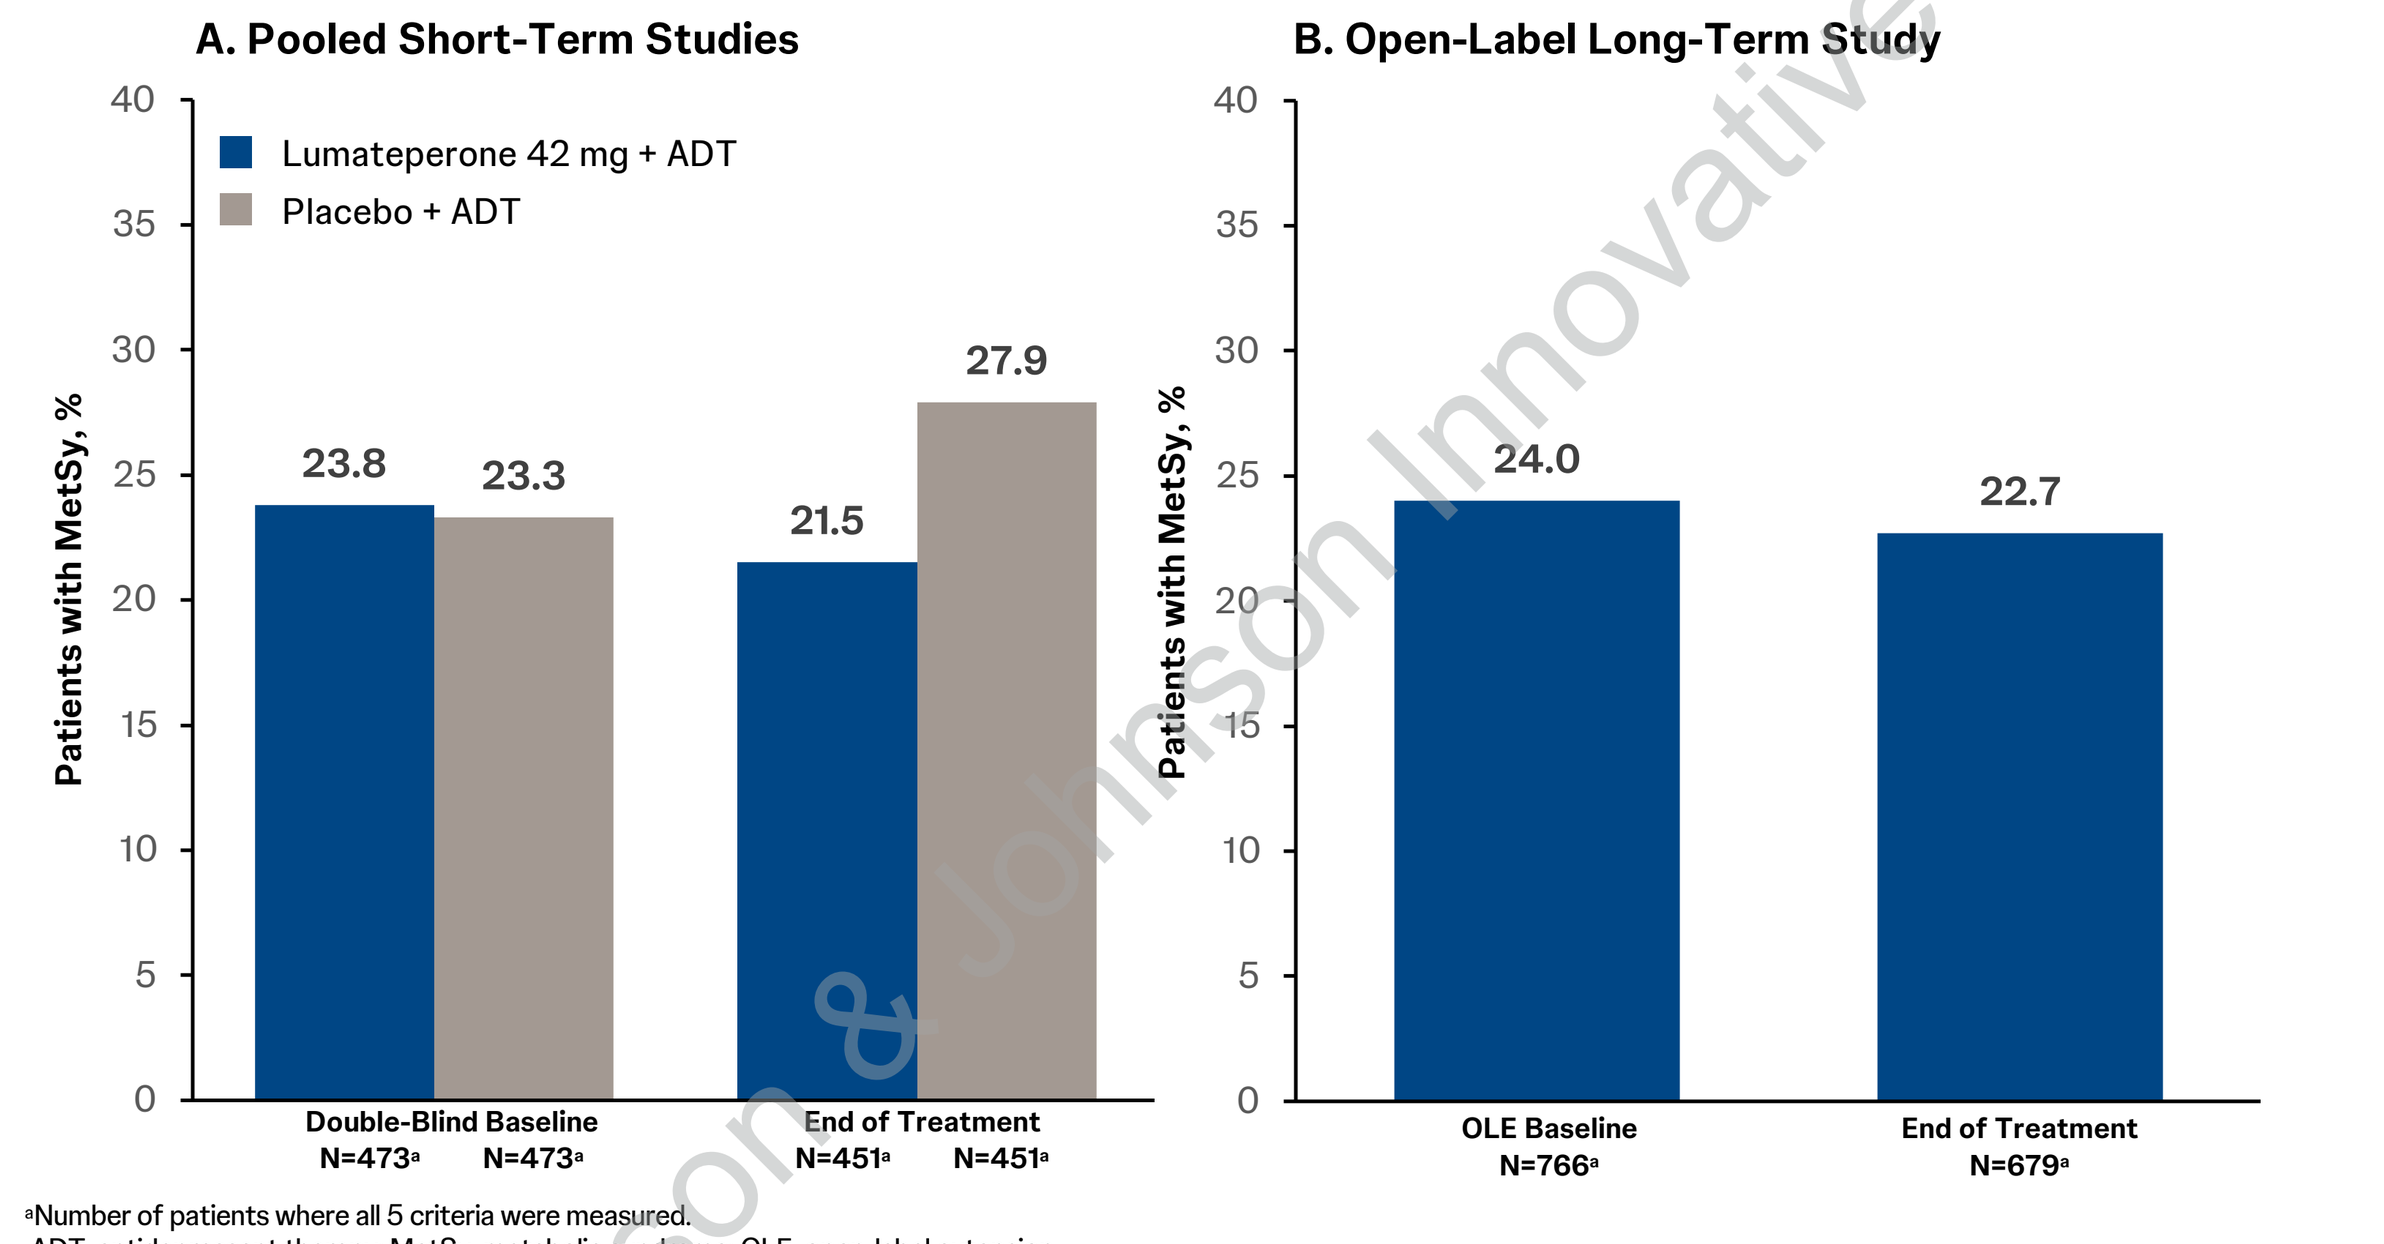
A. Pooled Short-Term Studies
B. Open-Label Long-Term Study
Lumateperone 42 mg + ADT
Placebo + ADT
Patients with MetSy, %
23.8
23.3
21.5
27.9
Double-Blind Baseline N=473ᵃ N=473ᵃ
End of Treatment N=451ᵃ N=451ᵃ
24.0
22.7
OLE Baseline N=766ᵃ
End of Treatment N=679ᵃ

ᵃNumber of patients where all 5 criteria were measured.
ADT, antidepressant therapy; MetSy, metabolic syndrome; OLE, open-label extension.

- Conversion analysis in the short-term studies showed that a greater percentage of patients treated with lumateperone 42 mg + ADT vs placebo + ADT improved their MetSy status (ie, having MetSy at baseline to no longer having MetSy at EOT (**Figure 2A**)
- In the OLE, the percentage of patients who converted to meeting MetSy criteria at EOT and those no longer meeting MetSy criteria at EOT were consistent (**Figure 2B**); 72.1% of patients at OLE baseline and 62.5% at EOT remained MetSy-free through the OLE study period

Figure 2. Conversion in Metabolic Syndrome Status from Baseline to End of Treatment in Pooled Short-Term (A) and Long-Term (B) Studies: Overall Population

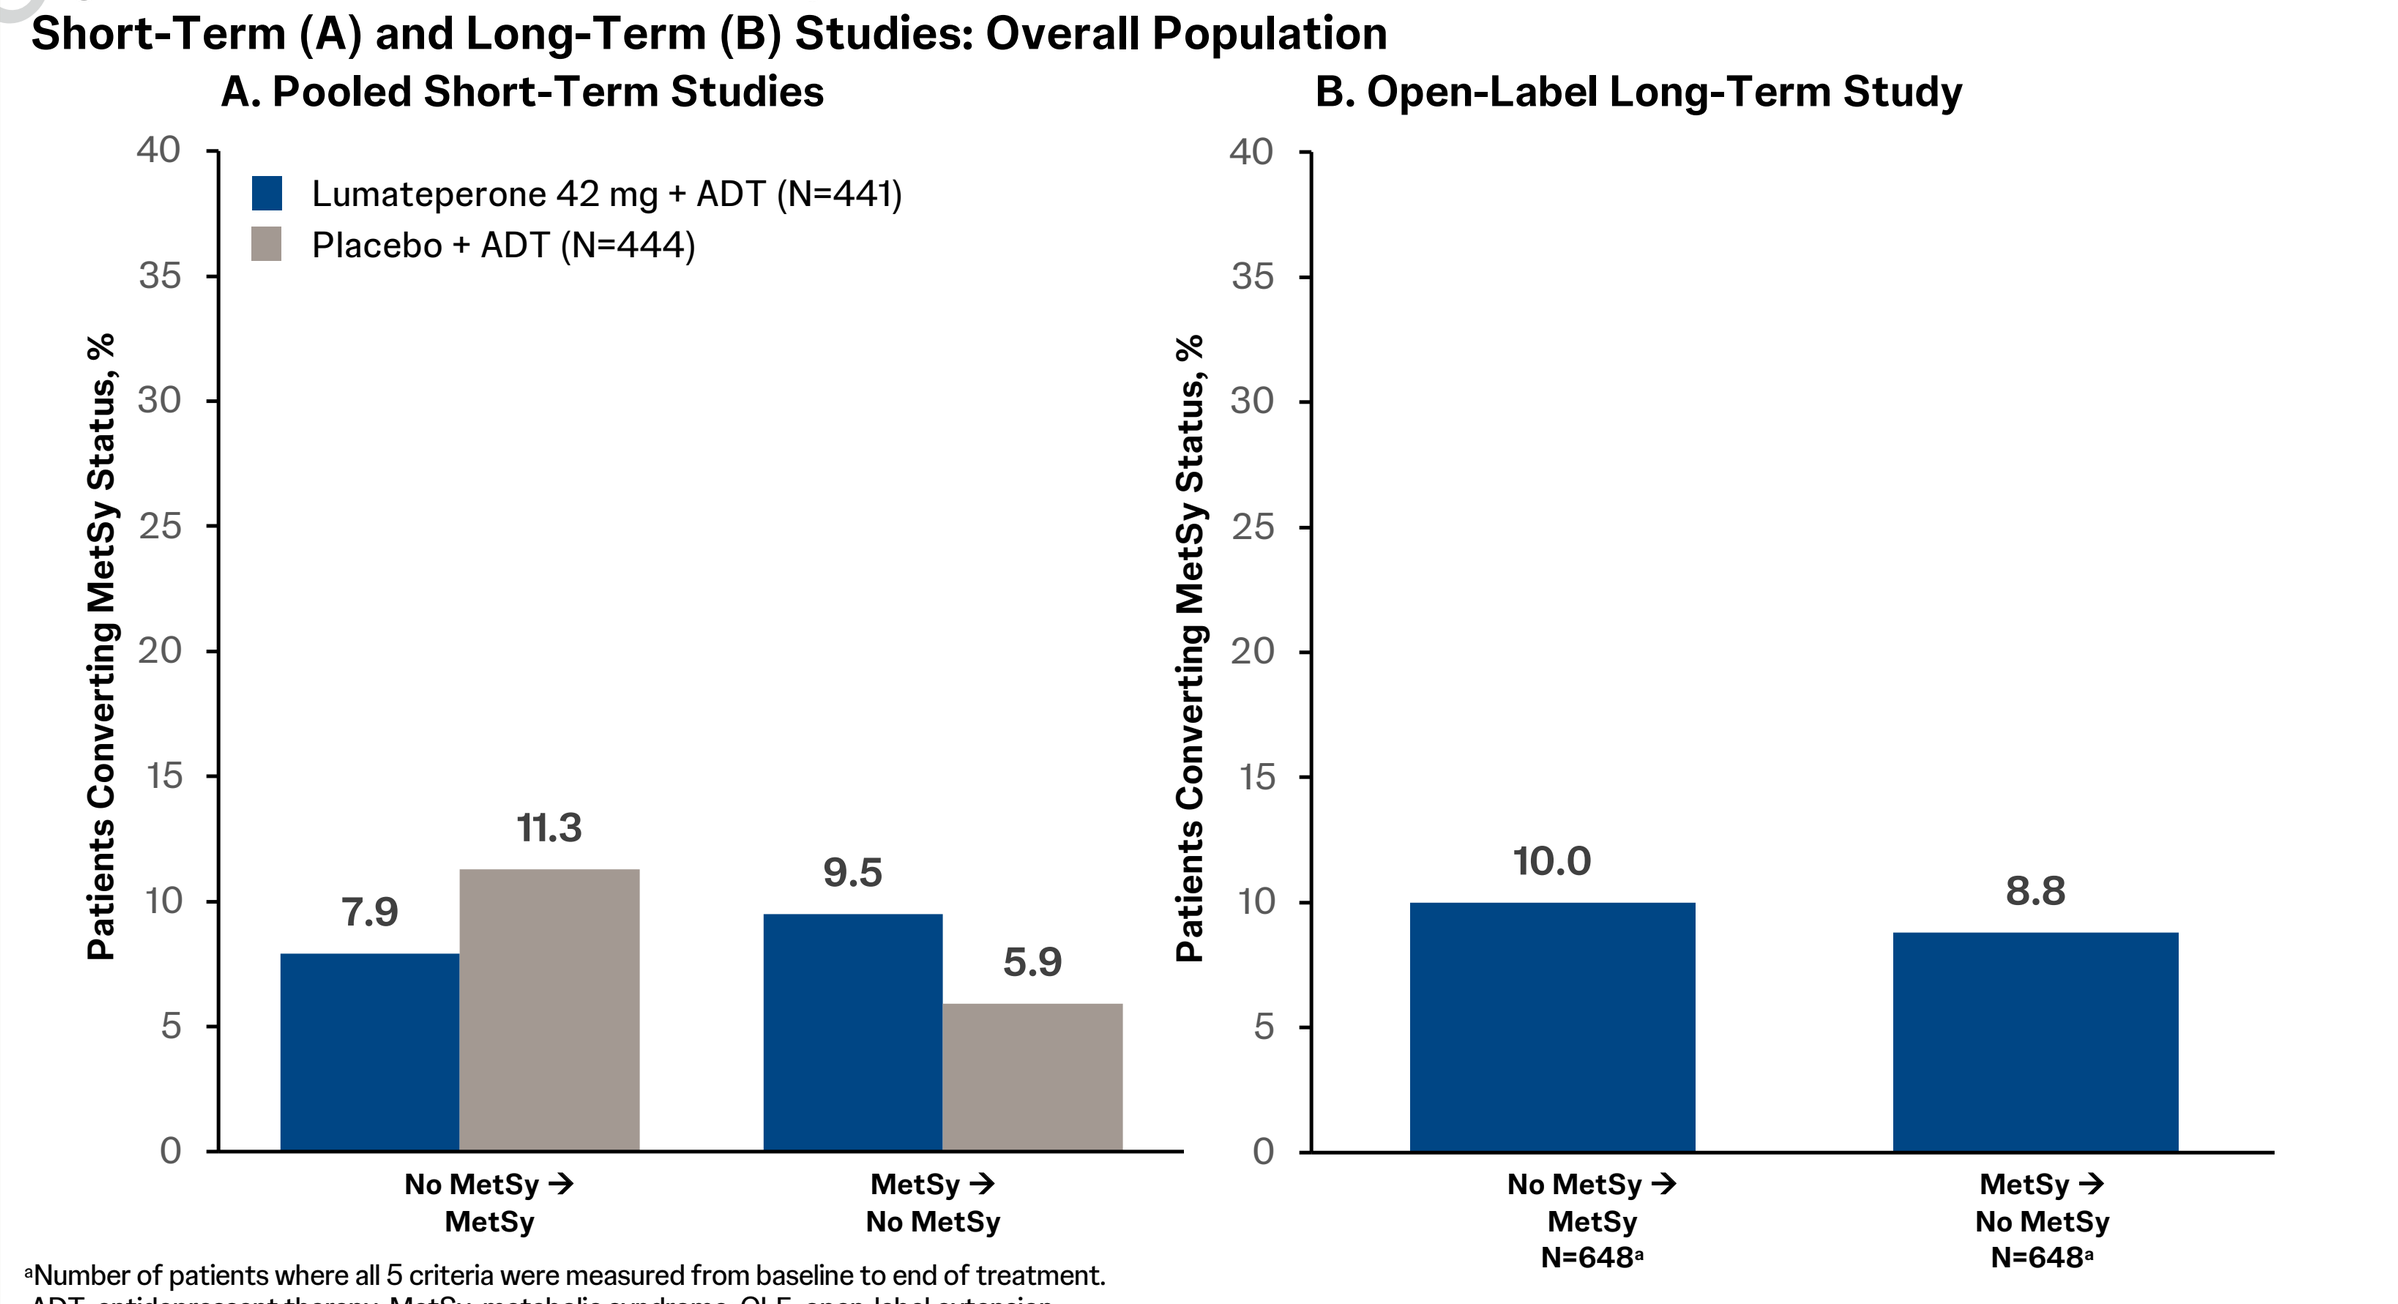
A. Pooled Short-Term Studies
B. Open-Label Long-Term Study
Lumateperone 42 mg + ADT (N=441)
Placebo + ADT (N=444)
Patients Converting MetSy Status, %
7.9
11.3
9.5
5.9
No MetSy → MetSy
MetSy → No MetSy
10.0
8.8
No MetSy → MetSy N=648ᵃ
MetSy → No MetSy N=648ᵃ

ᵃNumber of patients where all 5 criteria were measured from baseline to end of treatment.
ADT, antidepressant therapy; MetSy, metabolic syndrome; OLE, open-label extension.

Metabolic Syndrome: Concomitant ADT (SSRI vs SNRI)

- In the pooled short-term studies, the incidence of MetSy at EOT decreased from baseline with lumateperone 42 mg + concomitant SSRI, was consistent with lumateperone 42 mg + concomitant SNRI, and increased with placebo + ADT treatment (both concomitant SSRI and SNRI) (**Figure 3A**)
- Regardless of concomitant ADT taken, patients who received placebo + ADT were more likely to convert to having MetSy from baseline to EOT compared with those who received lumateperone 42 mg + ADT (**Figure 3B**)

Figure 3. Metabolic Syndrome at Baseline and End-of-Treatment (A) and Conversion in Metabolic Syndrome Status (B) in Pooled Short-Term Studies Stratified by ADT (SSRI vs SNRI)

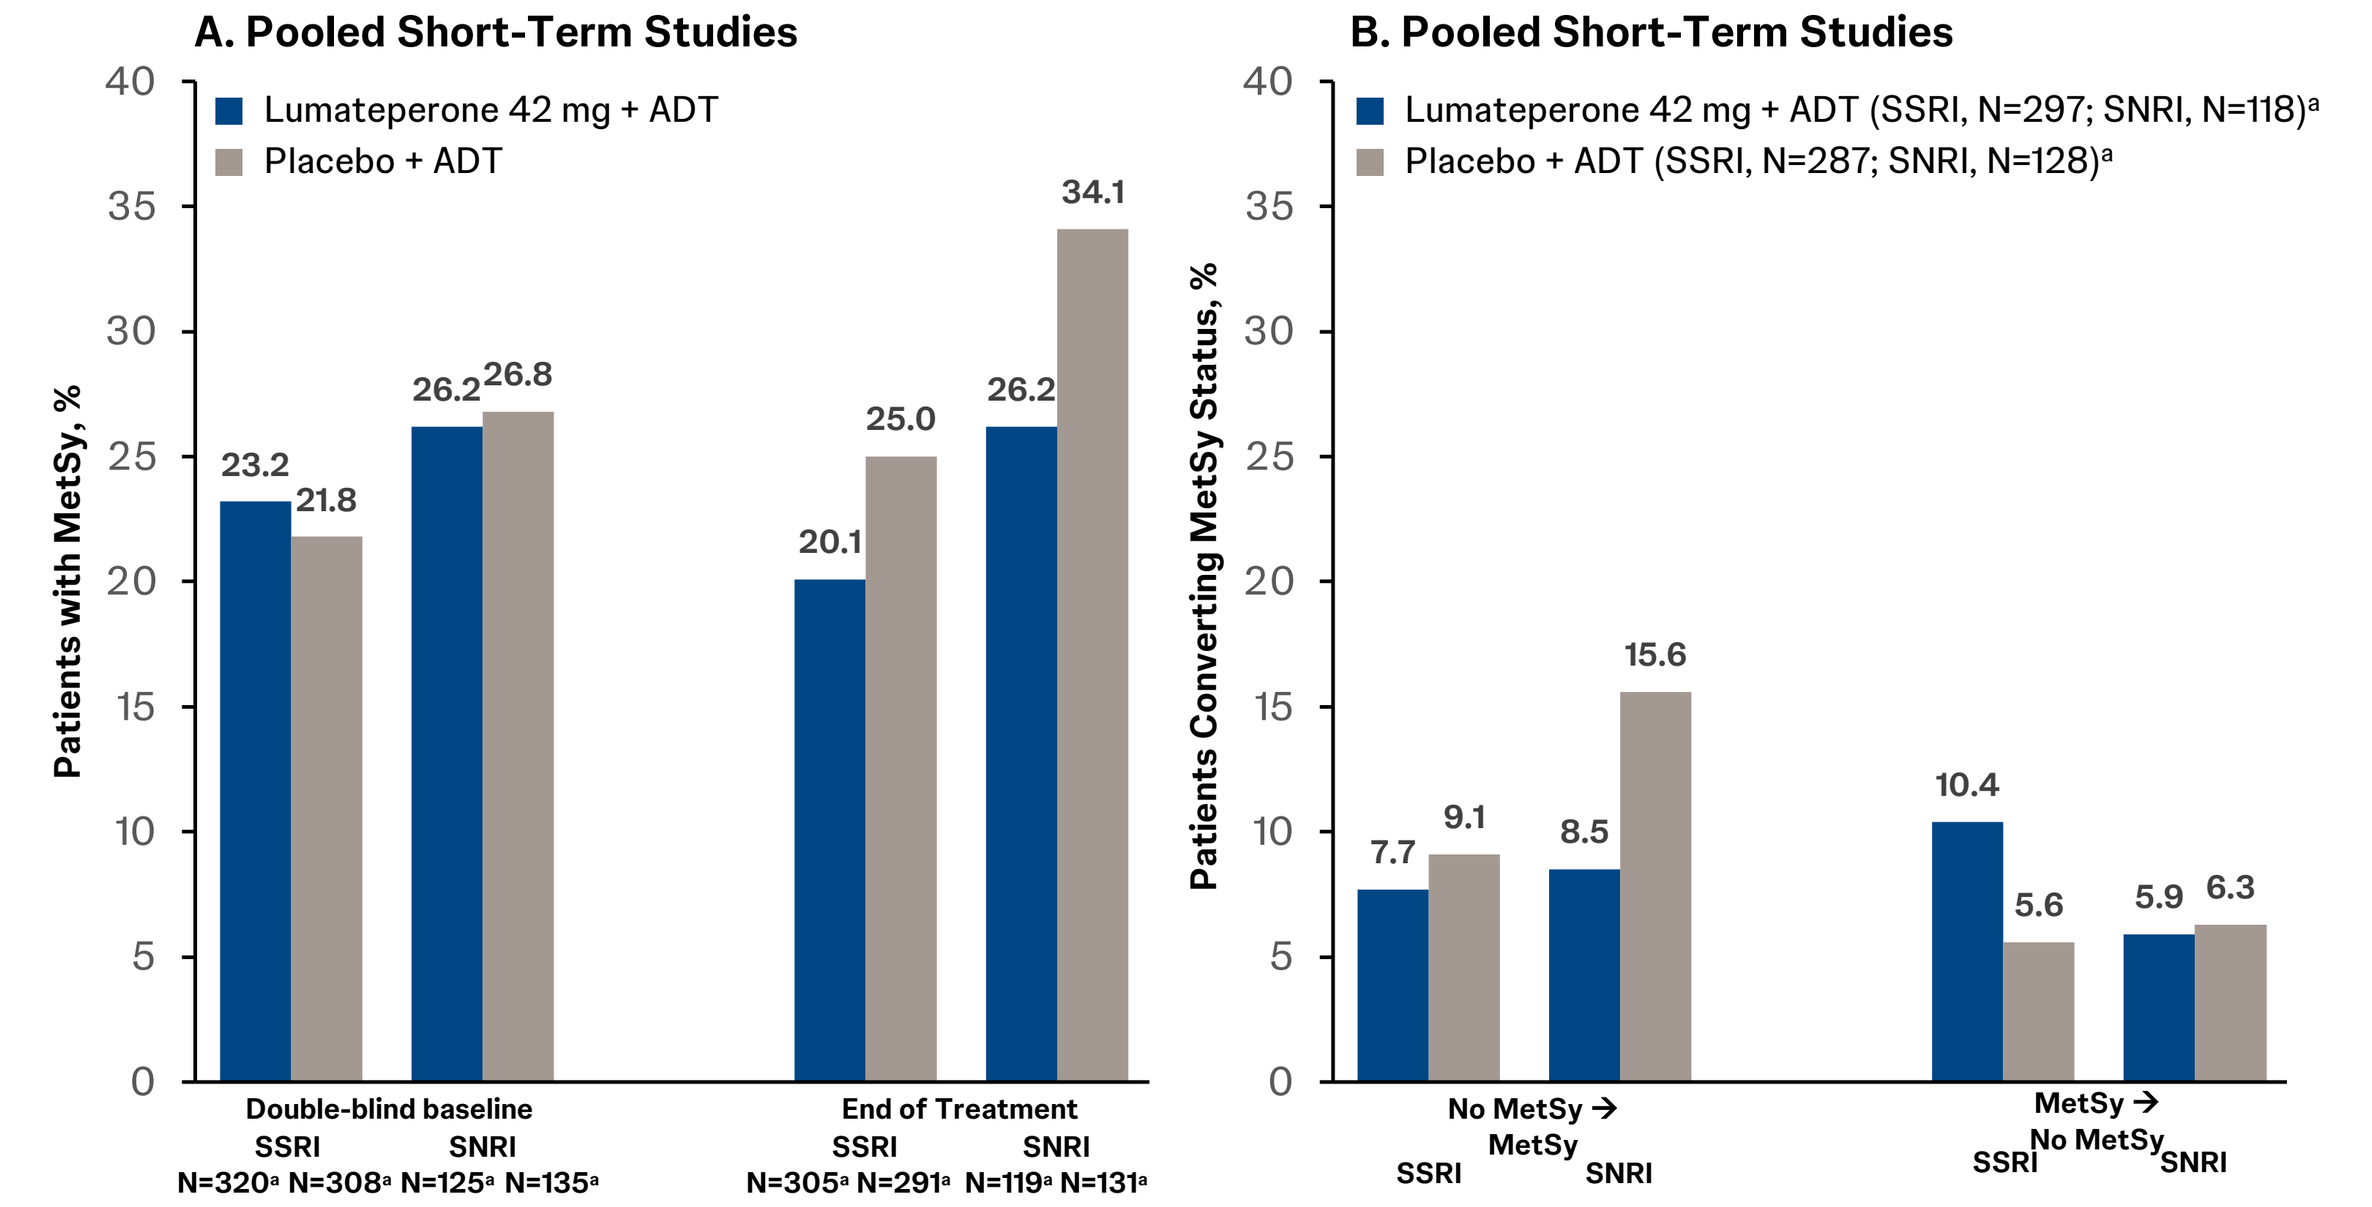
A. Pooled Short-Term Studies
B. Pooled Short-Term Studies
Lumateperone 42 mg + ADT
Placebo + ADT
Patients with MetSy, %
23.2
21.8
26.2
26.8
20.1
25.0
26.2
34.1
Double-blind baseline SSRI SNRI N=320ᵃ N=308ᵃ N=125ᵃ N=135ᵃ
End of Treatment SSRI SNRI N=305ᵃ N=291ᵃ N=119ᵃ N=131ᵃ
Lumateperone 42 mg + ADT (SSRI, N=297; SNRI, N=118)ᵃ
Placebo + ADT (SSRI, N=287; SNRI, N=128)ᵃ
Patients Converting MetSy Status, %
7.7
9.1
8.5
15.6
10.4
5.6
5.9
6.3
No MetSy → MetSy SSRI SNRI
MetSy → No MetSy SSRI SNRI

ᵃNumber of patients where all 5 criteria were measured.
ADT, antidepressant therapy; MetSy, metabolic syndrome; SNRI, serotonin-norepinephrine reuptake inhibitor; SSRI, selective serotonin reuptake inhibitor.

Metabolic Syndrome: Conversion in Status by Individual Criteria

- In the pooled short-term studies, the individual criteria that shifted the most from meeting MetSy criteria at baseline to no longer meeting criteria at EOT was systolic BP in the lumateperone 42 mg + ADT group (14.5%) and placebo + ADT group (14.6%), followed by triglycerides (10.6% vs 9.4%), fasting glucose (9.5% vs 6.3%), and HDL cholesterol (8.9% vs 7.3%)
- In the long-term study, the individual criteria that improved the most in terms of changing from meeting the MetSy criterion at baseline to no longer meeting it at EOT also included systolic BP

Hb$_{A1c}$ Status (≤7% vs >7%)

- In the OLE, 99.3% of patients at OLE baseline and 99.5% at EOT had Hb$_{A1c}$ ≤7%

Framingham Risk Scores

- Estimated 10-year cardiovascular disease (CVD) risk was calculated using the Framingham general CVD equation
- Mean Framingham risk (%) remained stable from the baseline of the double-blind randomized phase to EOT for lumateperone 42 mg + ADT compared with placebo + ADT (**Figure 4A**); results were consistent independent of smoking status
- In the OLE, mean Framingham risk scores were stable from OLE baseline to EOT (**Figure 4B**)

Figure 4. Mean 10-year Cardiovascular Risk by Framingham General CVD Equation in Pooled Short-Term (A) and Long-Term (B) Studies

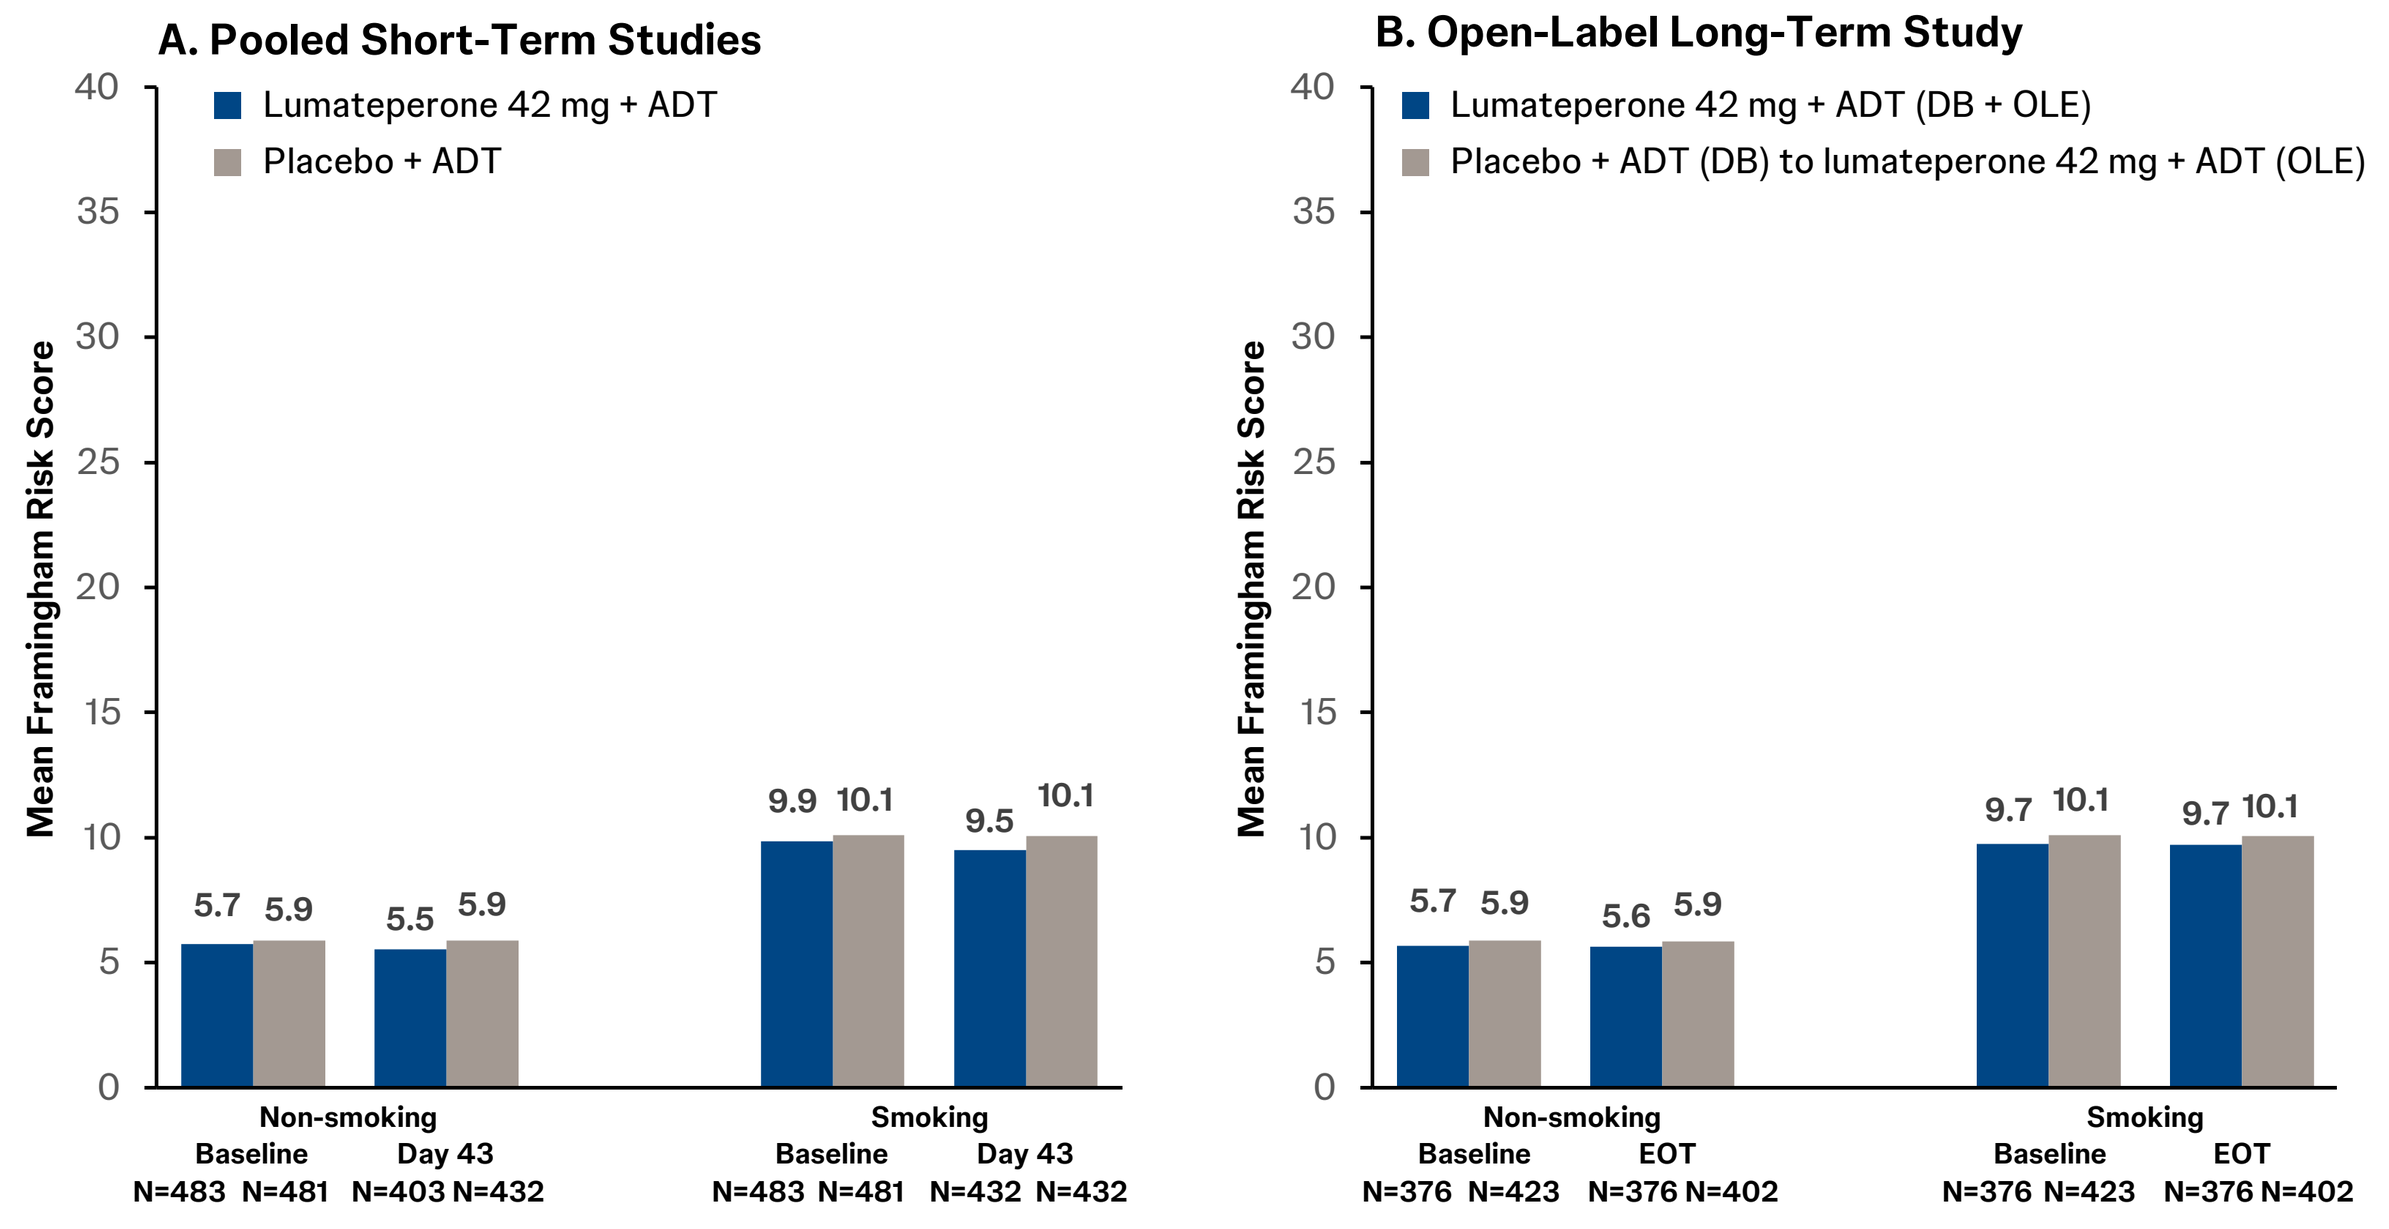
A. Pooled Short-Term Studies
Lumateperone 42 mg + ADT
Placebo + ADT
Mean Framingham Risk Score
5.7
5.9
5.5
5.9
9.9
10.1
9.5
10.1
Non-smoking Baseline Day 43 N=483 N=481 N=403 N=432
Smoking Baseline Day 43 N=483 N=481 N=432 N=432
B. Open-Label Long-Term Study
Lumateperone 42 mg + ADT (DB + OLE)
Placebo + ADT (DB) to lumateperone 42 mg + ADT (OLE)
5.7
5.9
5.6
5.9
9.7
10.1
9.7
10.1
Non-smoking Baseline EOT N=376 N=423 N=376 N=402
Smoking Baseline EOT N=376 N=423 N=376 N=402

ADT, antidepressant therapy; BP, blood pressure; CVD, cardiovascular disease; DB, double blind; HDL, high-density lipoprotein; MetSy, metabolic syndrome; OLE, open-label extension.

Conclusions

- In this post hoc analysis of pooled short-term studies and a long-term OLE study, the incidence of MetSy remained low in patients treated with lumateperone 42 mg + ADT treatment vs placebo + ADT
- A greater proportion of patients remained MetSy-free following lumateperone 42 mg + ADT treatment vs placebo + ADT treatment
- The incidence of MetSy-free patients was greater with lumateperone 42 mg + ADT than placebo + ADT with concomitant SSRI
- Lumateperone 42 mg + ADT did not increase estimated 10-year Framingham CVD risk compared with placebo + ADT over the double-blind treatment period; Framingham risk estimates remained stable in the OLE
- Long-term cardiovascular outcomes in patients treated long-term with lumateperone require additional investigation

Acknowledgments

The authors thank all study investigators, research staff, and patients for their participation. Medical writing support was provided by Apurva H. Davé, PhD, of Nucleus Global, an Inizio company, funded by Intra-Cellular Therapies, a Johnson & Johnson company.

Disclosures

This study was sponsored by Johnson & Johnson.

A Krystal has received research grants from Alkermes, Janssen Pharmaceuticals, Axsome Pharmaceuticals, Attune, Eisai, Harmony, Neumora, Neurocrine Biosciences, The Ray and Dagmar Dolby Family Fund, Weill Institute for Neurosciences, and the National Institutes of Health. He has consulted for Axsome Therapeutics, AbbVie, Big Health, Eisai, Evecxia, Harmony Biosciences, Idorsia, Janssen Pharmaceuticals, Jazz Pharmaceuticals, Neurocrine Biosciences, Neumora, NeuraWell, Otsuka, Sage, and Takeda. He has stock options in NeuraWell, and Big Health.

CN Epperson receives research grant support from Sage Therapeutics and HealthRhythms. She is a member of the Executive Board for Parthenon Management Group, for which she receives no consulting fees or equity. She is a member of the Advisory Board for BabyScripts where she has equity, and EmbarkNeuro, from which she receives consulting fees.

I Turkoz, KR Dorsey, and Z Zhang are employees of Johnson & Johnson.

B Curcio is a contractor of Johnson & Johnson Innovative Medicine R&D.

CU Correll has been a consultant and/or advisor to or has received honoraria from: AbbVie, Alkermes, Allergan, Angelini, Aristo, Autobahn, Axsome, Boehringer Ingelheim, Bristol Myers Squibb, Cardio Diagnostics, Cerevel, CNX Therapeutics, Compass Pathways, Darnytsia, Delpor, Denovo, Draig, Eli Lilly, Eumentis Therapeutics, Gedeon Richter, GH Research, Hetero, Hikma, Holmusk, IntraCellular Therapies, Jamjoom Pharma, Janssen/J&J, Karuna, LB Pharma, Lundbeck, MedInCell, MedLink Global, Merck, Mindpax, Mitsubishi Tanabe Pharmaceuticals, MapLight, Mylan, Neumora Therapeutics, Neuraxpharm, Neurocrine, Neurelis, Neurosterix, NeuShen, Neusignal Therapeutics, Newron, Noven, Novo Nordisk, Orion Pharma, Otsuka, PPD Biotech, Recognify Life Science, Recordati, Relmada, Response Pharmaceutical, Reviva, Rovi, Saladax, Sanofi, Seqirus, Servier, Sumitomo Pharma America, Sunovion, Sun Pharma, Supernus, Tabuk, Takeda, Teva, Terran, Tolmar, Vertex, Viatris, and Xenon Pharmaceuticals. He provided expert testimony for Janssen, Lundbeck, Neurocrine, and Otsuka. He served on a Data Safety Monitoring Board for Compass Pathways, IntraCellular Therapies, Relmada, Reviva, and Rovi. He has received grant support from Boehringer Ingelheim, Janssen, and Takeda. He received royalties from UpToDate and is also a stock option or stockholder of Cardio Diagnostics, Kuleon Biosciences, LB Pharmaceuticals, Medincell, MedLink Global, Mindpax, Quantic, and Terran.

Neuropsychiatry

Scan the QR code
The QR code is intended to provide scientific information for individual reference, and the information should not be altered or reproduced in any way

Presented at Psych Congress 2026, September 15-19, 2026, New Orleans, LA, USA

References:
1. Earley WR, et al. *CNS Drugs*. 2026; doi:10.1007/s40263-026-01314-8; 2. Berk M, et al. *World Psychiatry*. 2023;22:366-387; 3. Beilby J, et al. *Clin Biochem Rev*. 2004; 25(3):195-198; 4. Vancampfort D, et al. *World Psychiatry*. 2015;14:339-347; 5. Caplyta (lumateperone). Prescribing information. Intra-Cellular Therapies, Inc; 2026; 6. Davis RE, Correll CU. *Expert Rev Neurother*. 2016;16:601-614; 7. Sowa-Kucma M, et al. *Int J Mol Sci*. 2024;25:13289; 8. Durgam S, et al. *J Clin Psychiatry*. 2025;86(4):25m15848; 9. Durgam S, et al. *Am J Psychiatry*. 2025;182(12):1072-1082; 10. Durgam S, et al. *Eur Neuropsychopharmacol*. 2026; 108:112786; 11. Correll CU, et al. *World Psychiatry*. 2015;14:119-136; 12. De Hert M, et al. *Nat Rev Endocrinol*. 2012;8, 114-126; 13. D'Agostino RB, et al. *Circulation*. 2008;117:743-753.

Email questions to Christoph U. Correll at christophcorrell@hotmail.com