100

Metabolic Profile of Adjunctive Lumateperone 42 mg in Major Depressive Disorder: A Pooled Analysis of 2 Randomized Placebo-Controlled Trials

Craig Chepke, MD[1]; Willie R. Earley, MD[2]; Susan G. Kozauer, MD[3]; Raffaele Migliore, MA[2]; Samar Madi, PhD[2]

[1]Excel Psychiatric Associates, Huntersville, NC, USA; [2]Intra-Cellular Therapies, a Johnson & Johnson company, Bedminster, NJ, USA; [3]Former employee of Intra-Cellular Therapies, a Johnson & Johnson company, Bedminster, NJ, USA

Background

- Major depressive disorder (MDD) is a highly prevalent and disabling condition characterized by persistent low mood, anhedonia, and cognitive and physical symptoms that impair daily functioning[1-3]
- Importantly, MDD is frequently associated with comorbidities, including cardiovascular and metabolic disorders[4-6]
- First-line treatment for MDD typically includes antidepressant therapy (ADT), such as a selective serotonin reuptake inhibitor or serotonin-norepinephrine reuptake inhibitor; however, currently available treatments are associated with weight gain and metabolic disturbances[7,8]
- Approximately 50% of patients with MDD have an inadequate ADT response, defined as not achieving ≥50% improvement in depression severity after ≥6-8 weeks of treatment[9]
- Although augmenting ADTs with atypical antipsychotics has received increased attention in recent years, this treatment regimen has been associated with safety concerns, including extrapyramidal symptoms, metabolic effects, and weight gain[10-12]
- Lumateperone is an atypical antipsychotic indicated for the treatment of MDD in adults as an adjunct to ADT; the treatment of schizophrenia in adults; and the treatment of depressive episodes associated with bipolar I or II disorder (bipolar depression) in adults, as monotherapy and as adjunctive therapy with lithium or valproate[13]
- Two Phase 3, randomized, double-blind, placebo-controlled studies (Study 501 [NCT04985942] and Study 502 [NCT05061706]) demonstrated the efficacy and safety of lumateperone 42 mg + ADT in patients with MDD with an inadequate ADT response[14,15]
 - In both trials, lumateperone 42 mg + ADT met primary and key secondary efficacy endpoints, significantly improving Montgomery-Åsberg Depression Rating Scale (MADRS) Total score and Clinical Global Impression–Severity (CGI-S) score from baseline to Day 43 compared with placebo + ADT
- A pooled analysis of these studies evaluated the metabolic profile associated with lumateperone 42 mg adjunctive to ADT

Methods

- In this post hoc analysis, safety data were pooled from Studies 501 and 502, both of which evaluated oral lumateperone 42 mg + ADT vs placebo + ADT in eligible adults (aged 18-65 years) who met the *Diagnostic and Statistical Manual of Mental Disorders, Fifth Edition* criteria for MDD with an inadequate response to 1-2 ADTs in the current depressive episode[14,15]
- Patients with MADRS Total score ≥24, CGI-S score ≥4, and Quick Inventory of Depressive Symptomatology-Self Report-16 item score ≥14 were randomized to 6-week lumateperone 42 mg + ADT or placebo + ADT[14,15]
- Assessments included changes in body mass index (BMI), weight, waist circumference, cardiometabolic laboratory parameters (glucose, cholesterol [total, high-density lipoprotein, low-density lipoprotein], triglycerides, insulin), and prolactin levels
 - Subgroups were analyzed by baseline BMI category (normal weight: ≥18.5 to <25 kg/m²; overweight: ≥25 to <30 kg/m²; obese: ≥30 kg/m²)

Results

Overall Population

- The pooled safety population comprised 964 patients (lumateperone + ADT, n=483; placebo + ADT, n=481), and 91.4% completed treatment
- Demographics and baseline characteristics were similar between groups
- Changes from baseline to end of treatment (EOT) were minimal with lumateperone + ADT vs placebo + ADT for weight (−0.1 vs +0.0 kg), BMI (−0.0 vs +0.0 kg/m²), and waist circumference (−0.2 vs −0.3 cm)
- Potentially clinically significant (PCS) weight increase (≥7% from baseline) was rare during treatment (lumateperone + ADT, 0.4%; placebo + ADT, 1.3%)
- No clinically meaningful changes were observed in cardiometabolic parameters or prolactin levels

Shift in BMI Categories

- At baseline, the proportions of patients across BMI categories were similar: normal weight (lumateperone + ADT, 32.3% vs placebo + ADT, 34.3%), overweight (38.5% vs 31.8%), and obese (29.2% vs 33.9%) (**Figure 1**)
 - The proportion of patients across BMI subgroups at EOT was also similar between lumateperone + ADT and placebo + ADT arms

Figure 1. Proportion of Patients in BMI Categories at Baseline and EOT

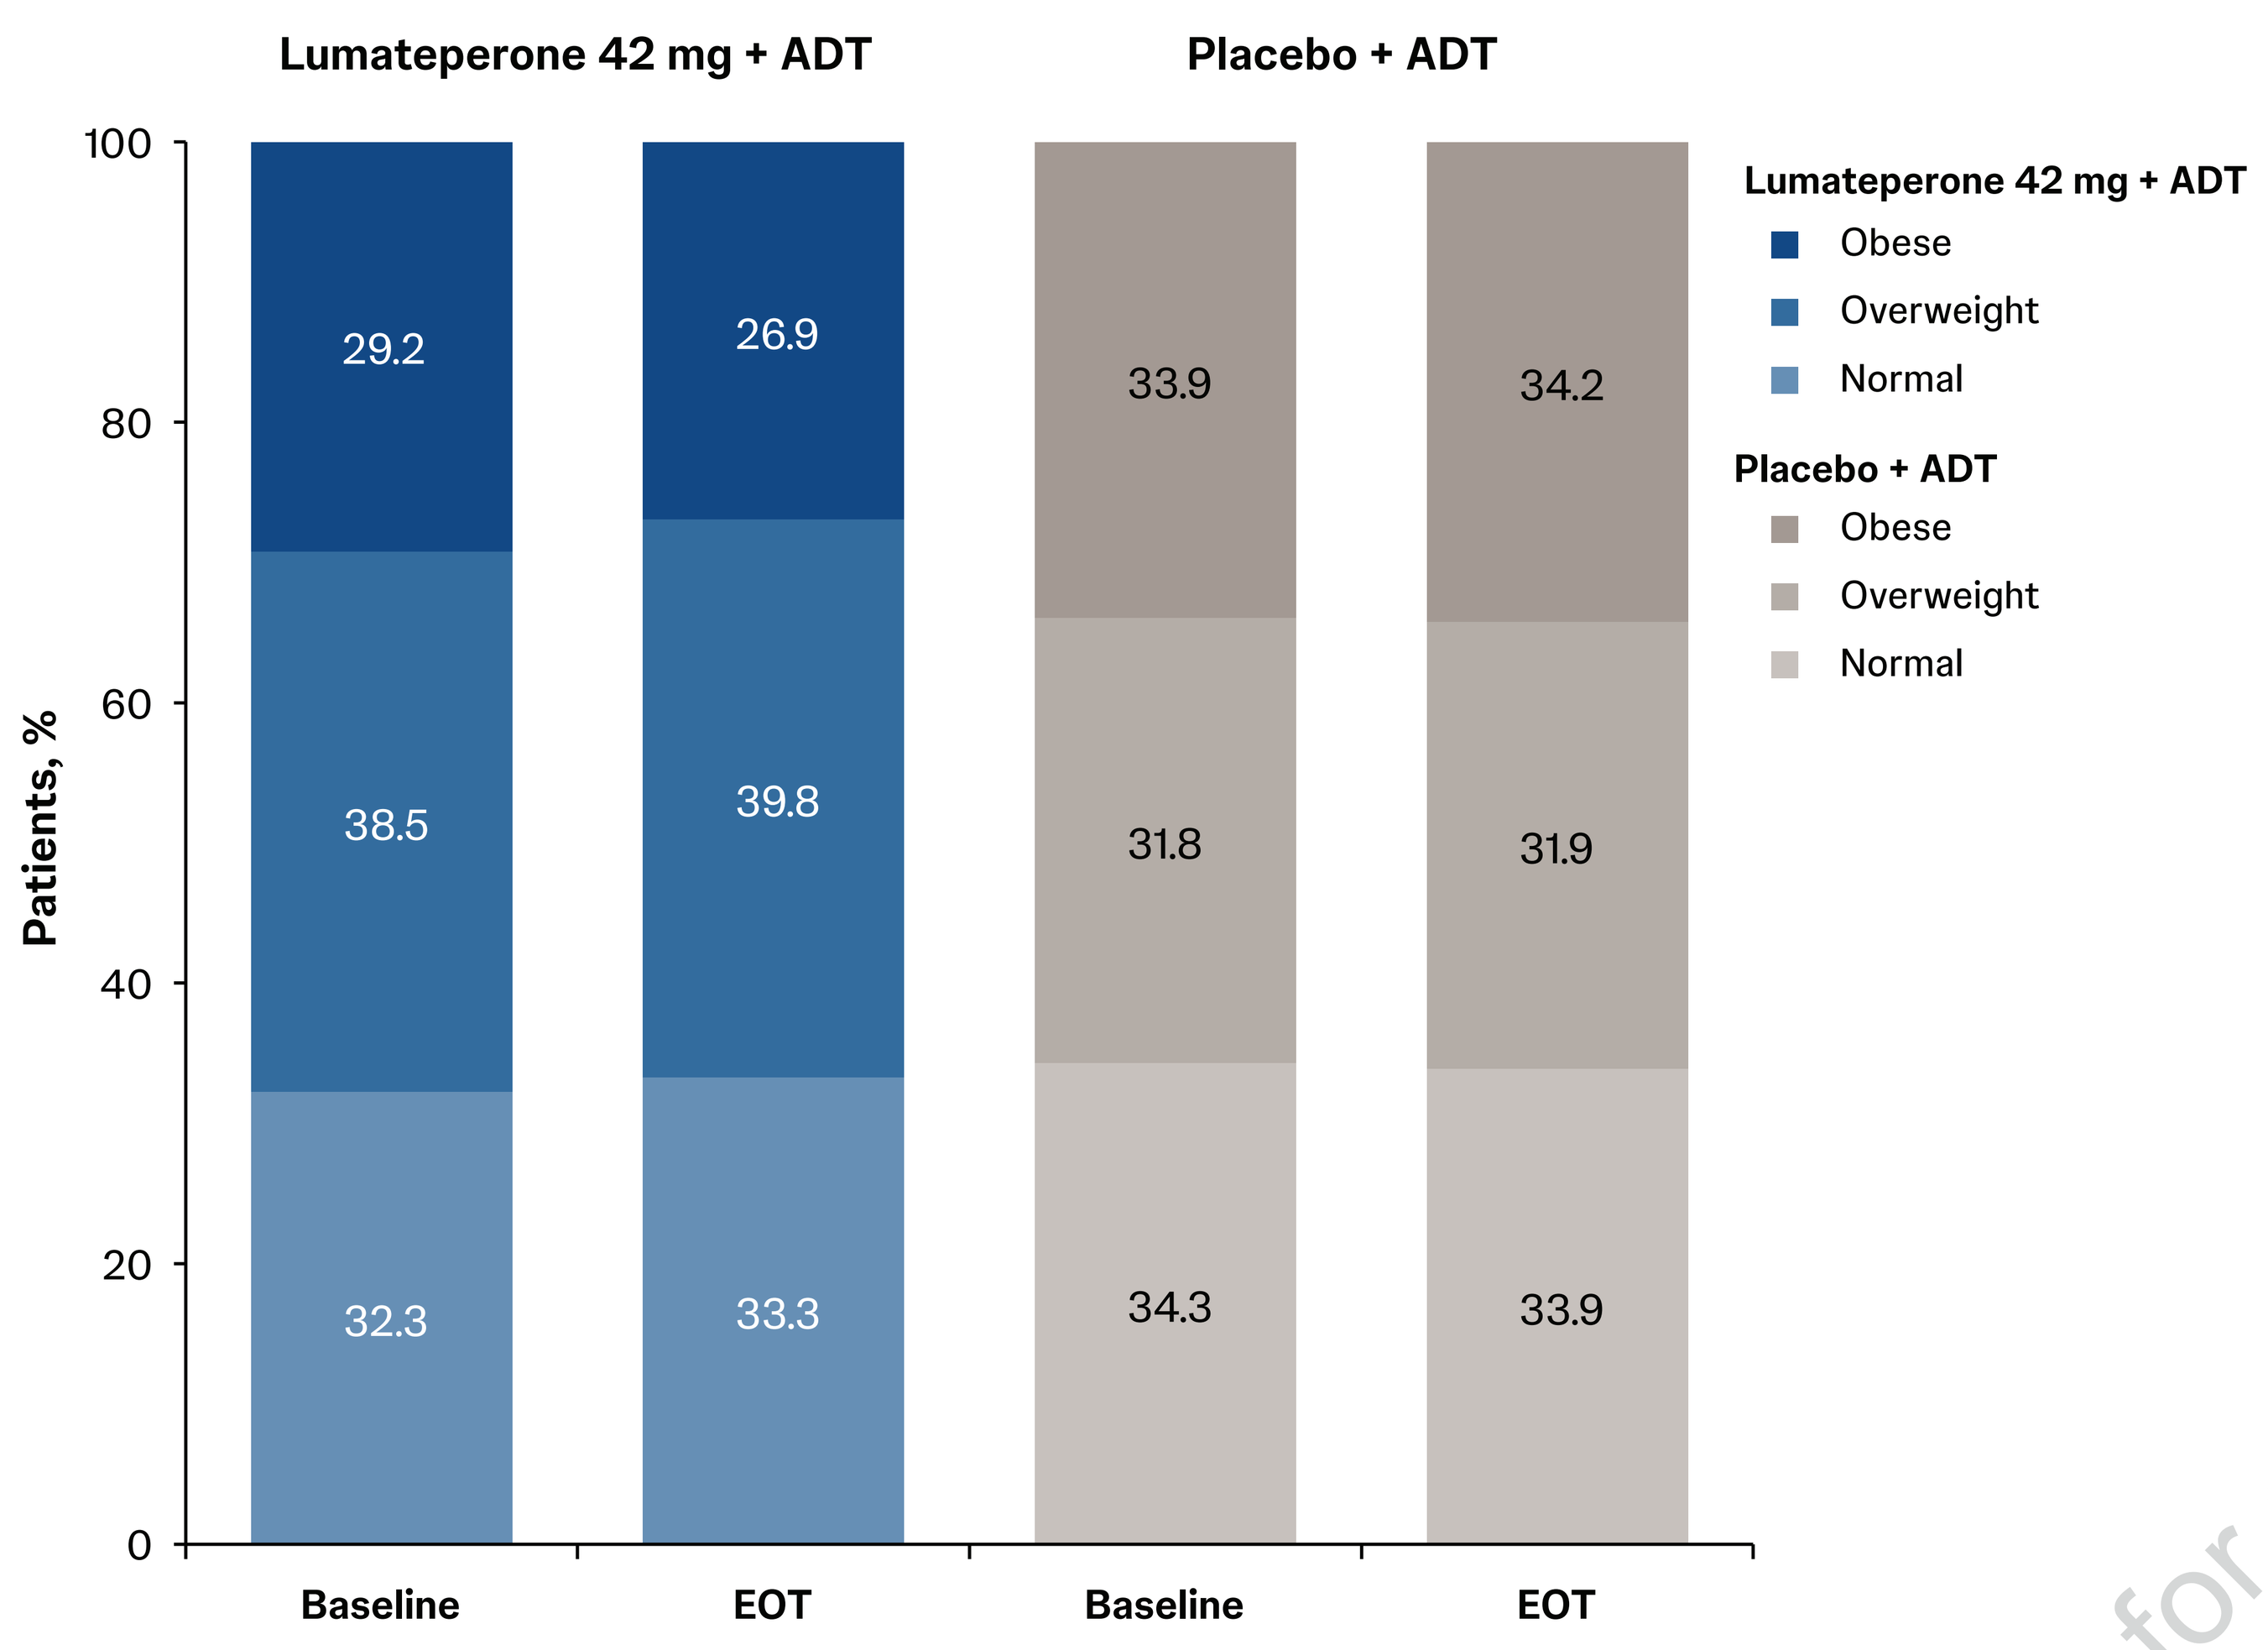
Lumateperone 42 mg + ADT
Placebo + ADT
Patients, %
100
80
60
40
20
0
29.2
38.5
32.3
26.9
39.8
33.3
33.9
31.8
34.3
34.2
31.9
33.9
Baseline
EOT
Baseline
EOT
Lumateperone 42 mg + ADT
Obese
Overweight
Normal
Placebo + ADT
Obese
Overweight
Normal

ADT, antidepressant therapy; BMI, body mass index; EOT, end of treatment.

- Most patients remained in their BMI category at EOT (normal weight: lumateperone + ADT, 91.0% vs placebo + ADT, 95.2%; overweight: 89.8% vs 86.9%; obese: 91.5% vs 94.5%)
 - Shifts in BMI categories from baseline to EOT were similar between the lumateperone + ADT and placebo + ADT groups (**Figure 2**)

Figure 2. Shifts in BMI Categories From Baseline to EOT

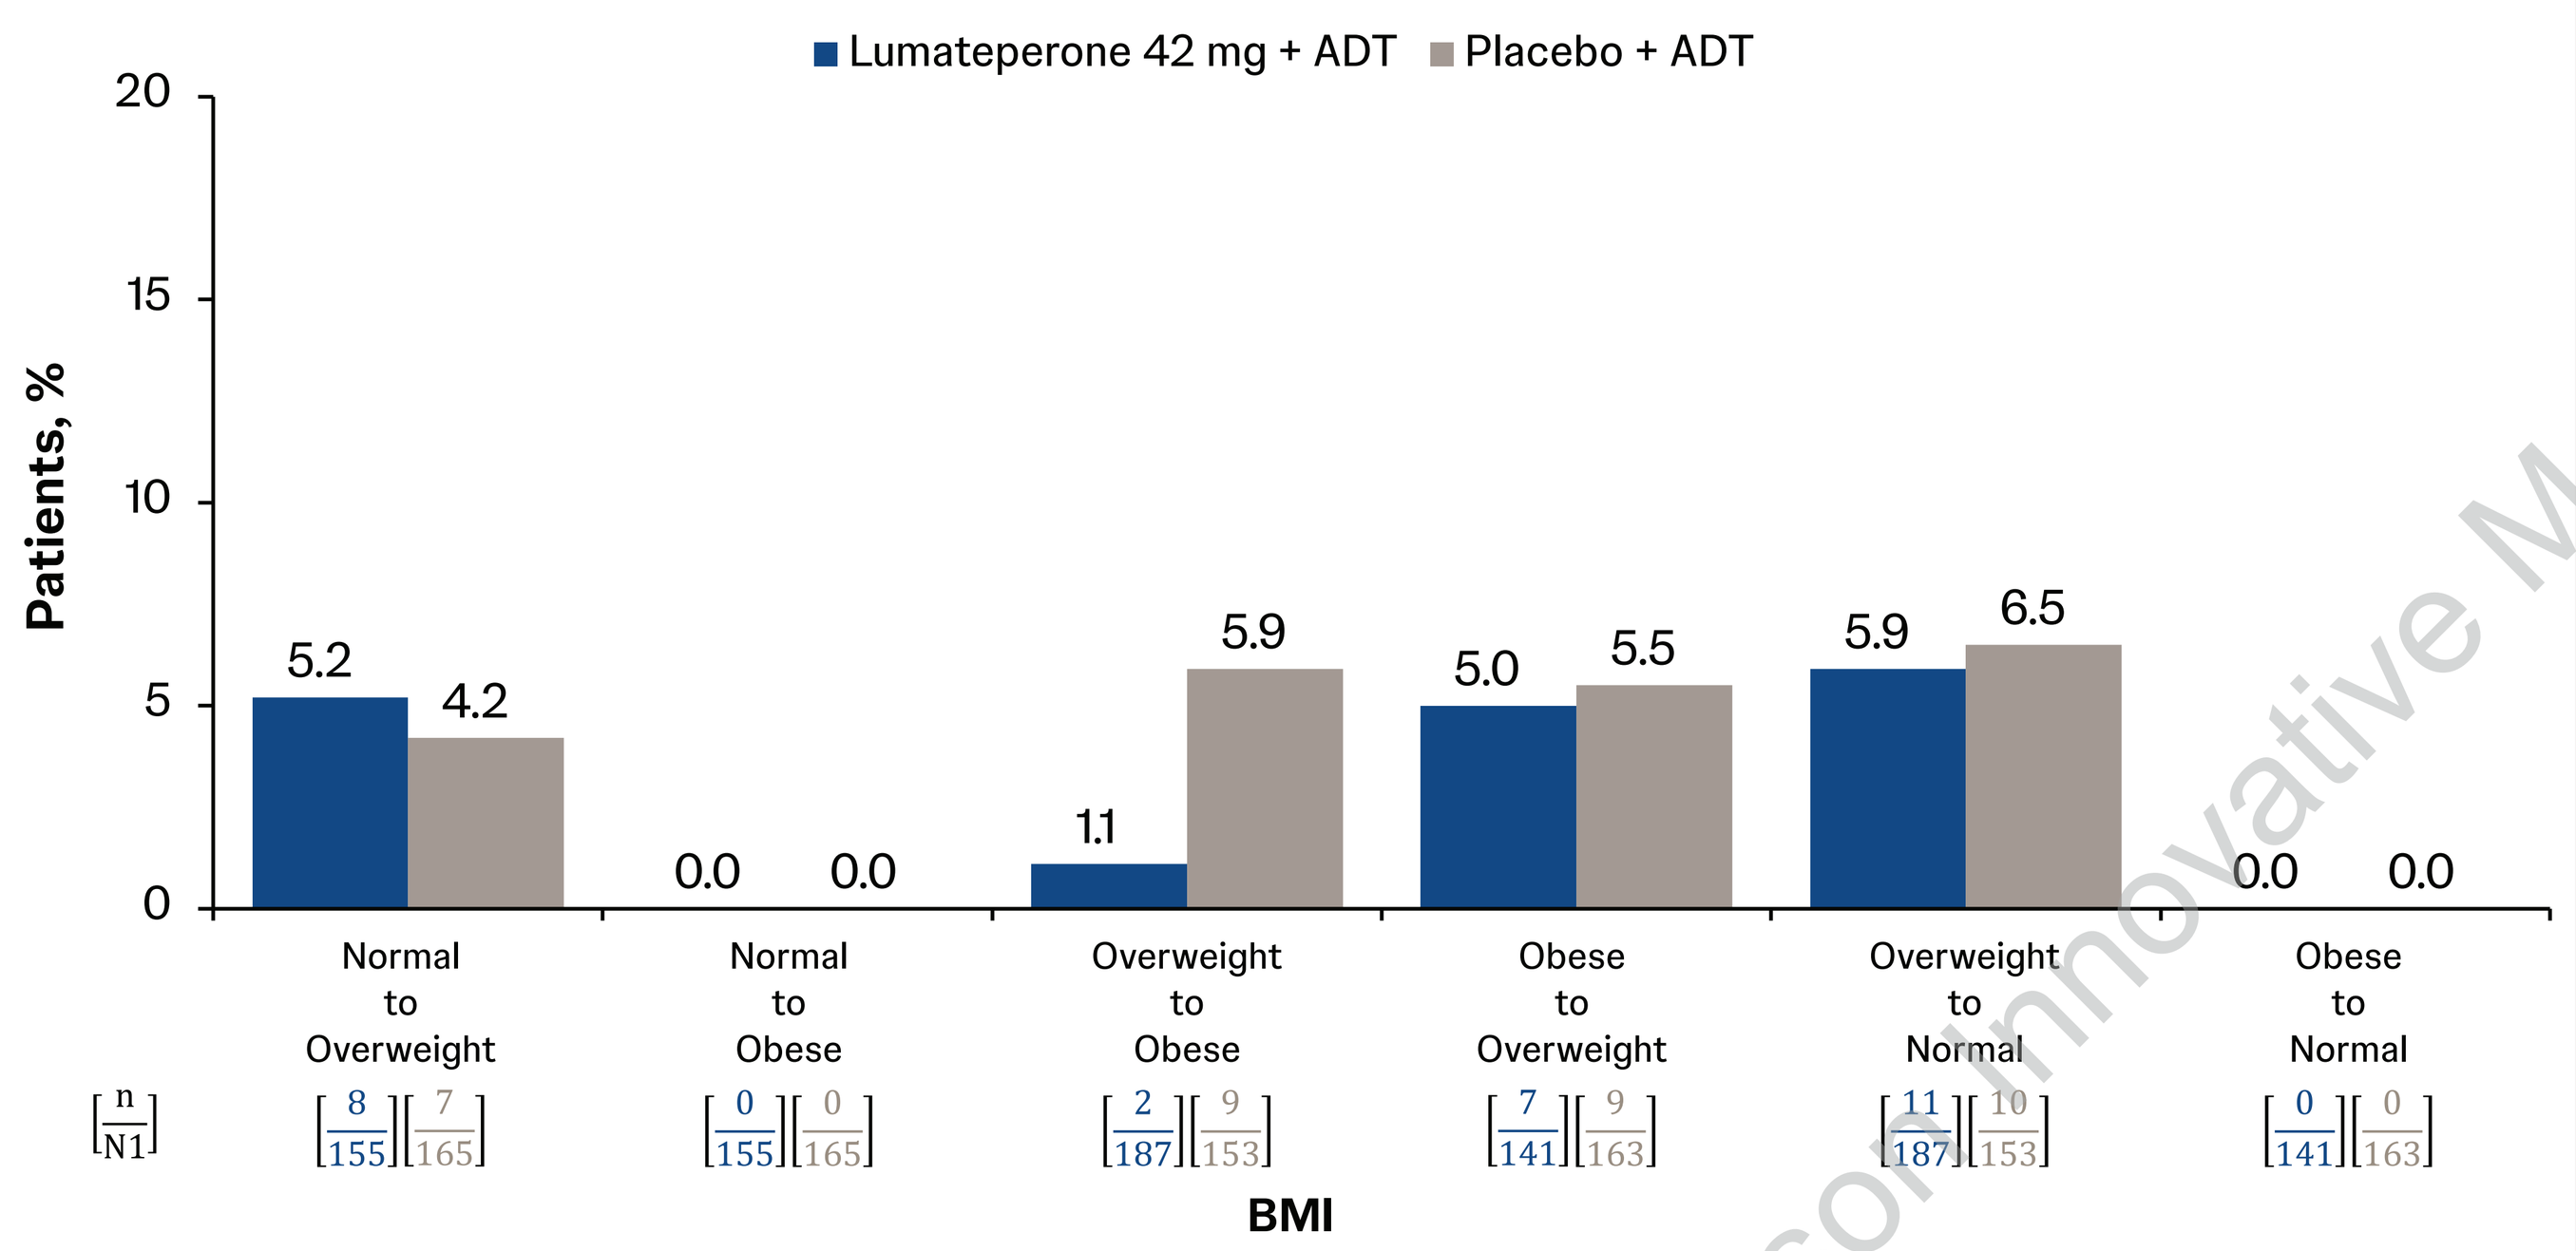
Lumateperone 42 mg + ADT
Placebo + ADT
Patients, %
20
15
10
5
0
5.2
4.2
0.0
0.0
1.1
5.9
5.0
5.5
5.9
6.5
0.0
0.0
Normal to Overweight
Normal to Obese
Overweight to Obese
Obese to Overweight
Overweight to Normal
Obese to Normal
n/N1
8/155 7/165
0/155 0/165
2/187 9/153
7/141 9/163
11/187 10/153
0/141 0/163
BMI

N1 = number of patients meeting the criteria at baseline.
Normal: ≥18.5 kg/m² and <25 kg/m²; Overweight: ≥25 kg/m² and <30 kg/m²; Obese: ≥30 kg/m².
BMI, body mass index; EOT, end of treatment.

Body Morphology Assessments

- Changes at EOT were minimal across BMI subgroups and treatment arms for BMI (−0.1 to +0.1 kg/m²), weight (−0.3 to +0.3 kg), and waist circumference (−0.6 to +0.1 cm) (**Figure 3**)
- PCS weight increase was rare with lumateperone + ADT vs placebo + ADT (normal weight: 0.7% vs 1.2%; overweight: 0.6% vs 0%; obese: 0% vs 2.5%) (**Figure 4A**)
 - PCS weight decrease (≥7%) was also rare during treatment (**Figure 4B**)

Figure 3. Changes in BMI, Weight, and Waist Circumference by Baseline BMI Category

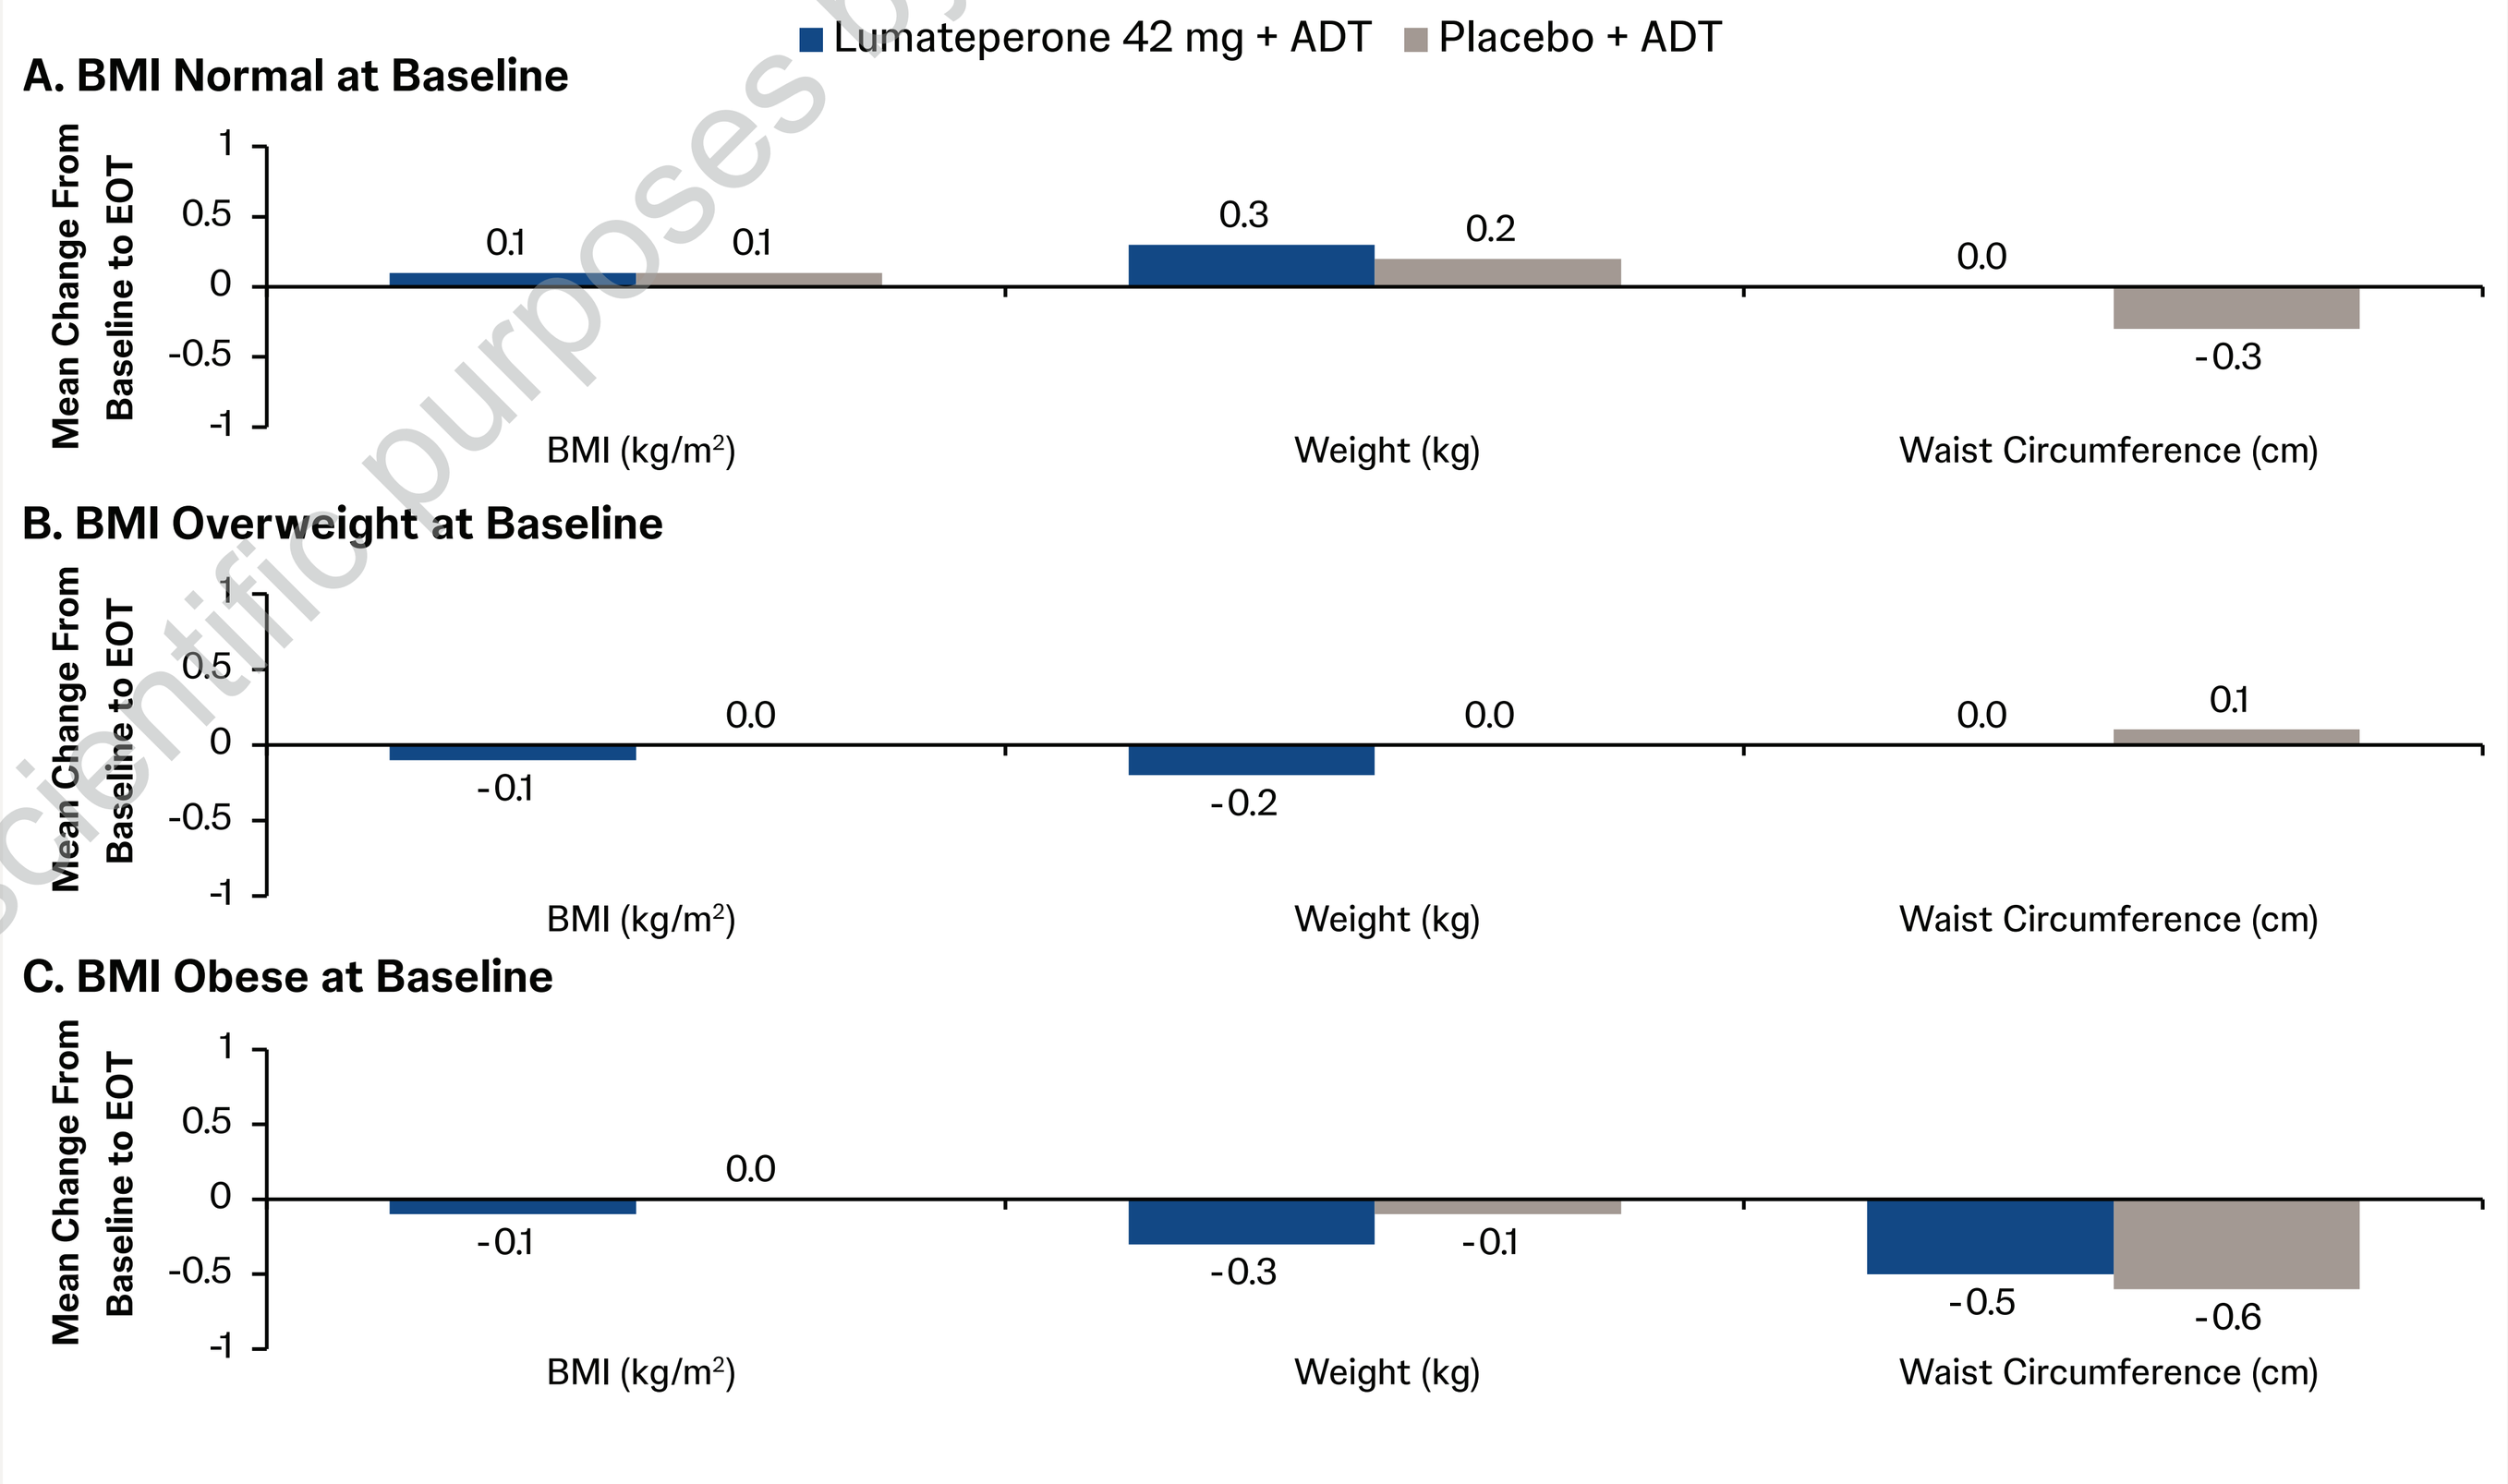
Lumateperone 42 mg + ADT
Placebo + ADT
A. BMI Normal at Baseline
Mean Change From Baseline to EOT
0.1
0.1
0.3
0.2
0.0
-0.3
BMI (kg/m²)
Weight (kg)
Waist Circumference (cm)
B. BMI Overweight at Baseline
-0.1
0.0
-0.2
0.0
0.0
0.1
BMI (kg/m²)
Weight (kg)
Waist Circumference (cm)
C. BMI Obese at Baseline
-0.1
0.0
-0.3
-0.1
-0.5
-0.6
BMI (kg/m²)
Weight (kg)
Waist Circumference (cm)

ADT, antidepressant therapy; BMI, body mass index; EOT, end of treatment.

Figure 4. Incidence of PCS Weight Changes by Baseline BMI Category

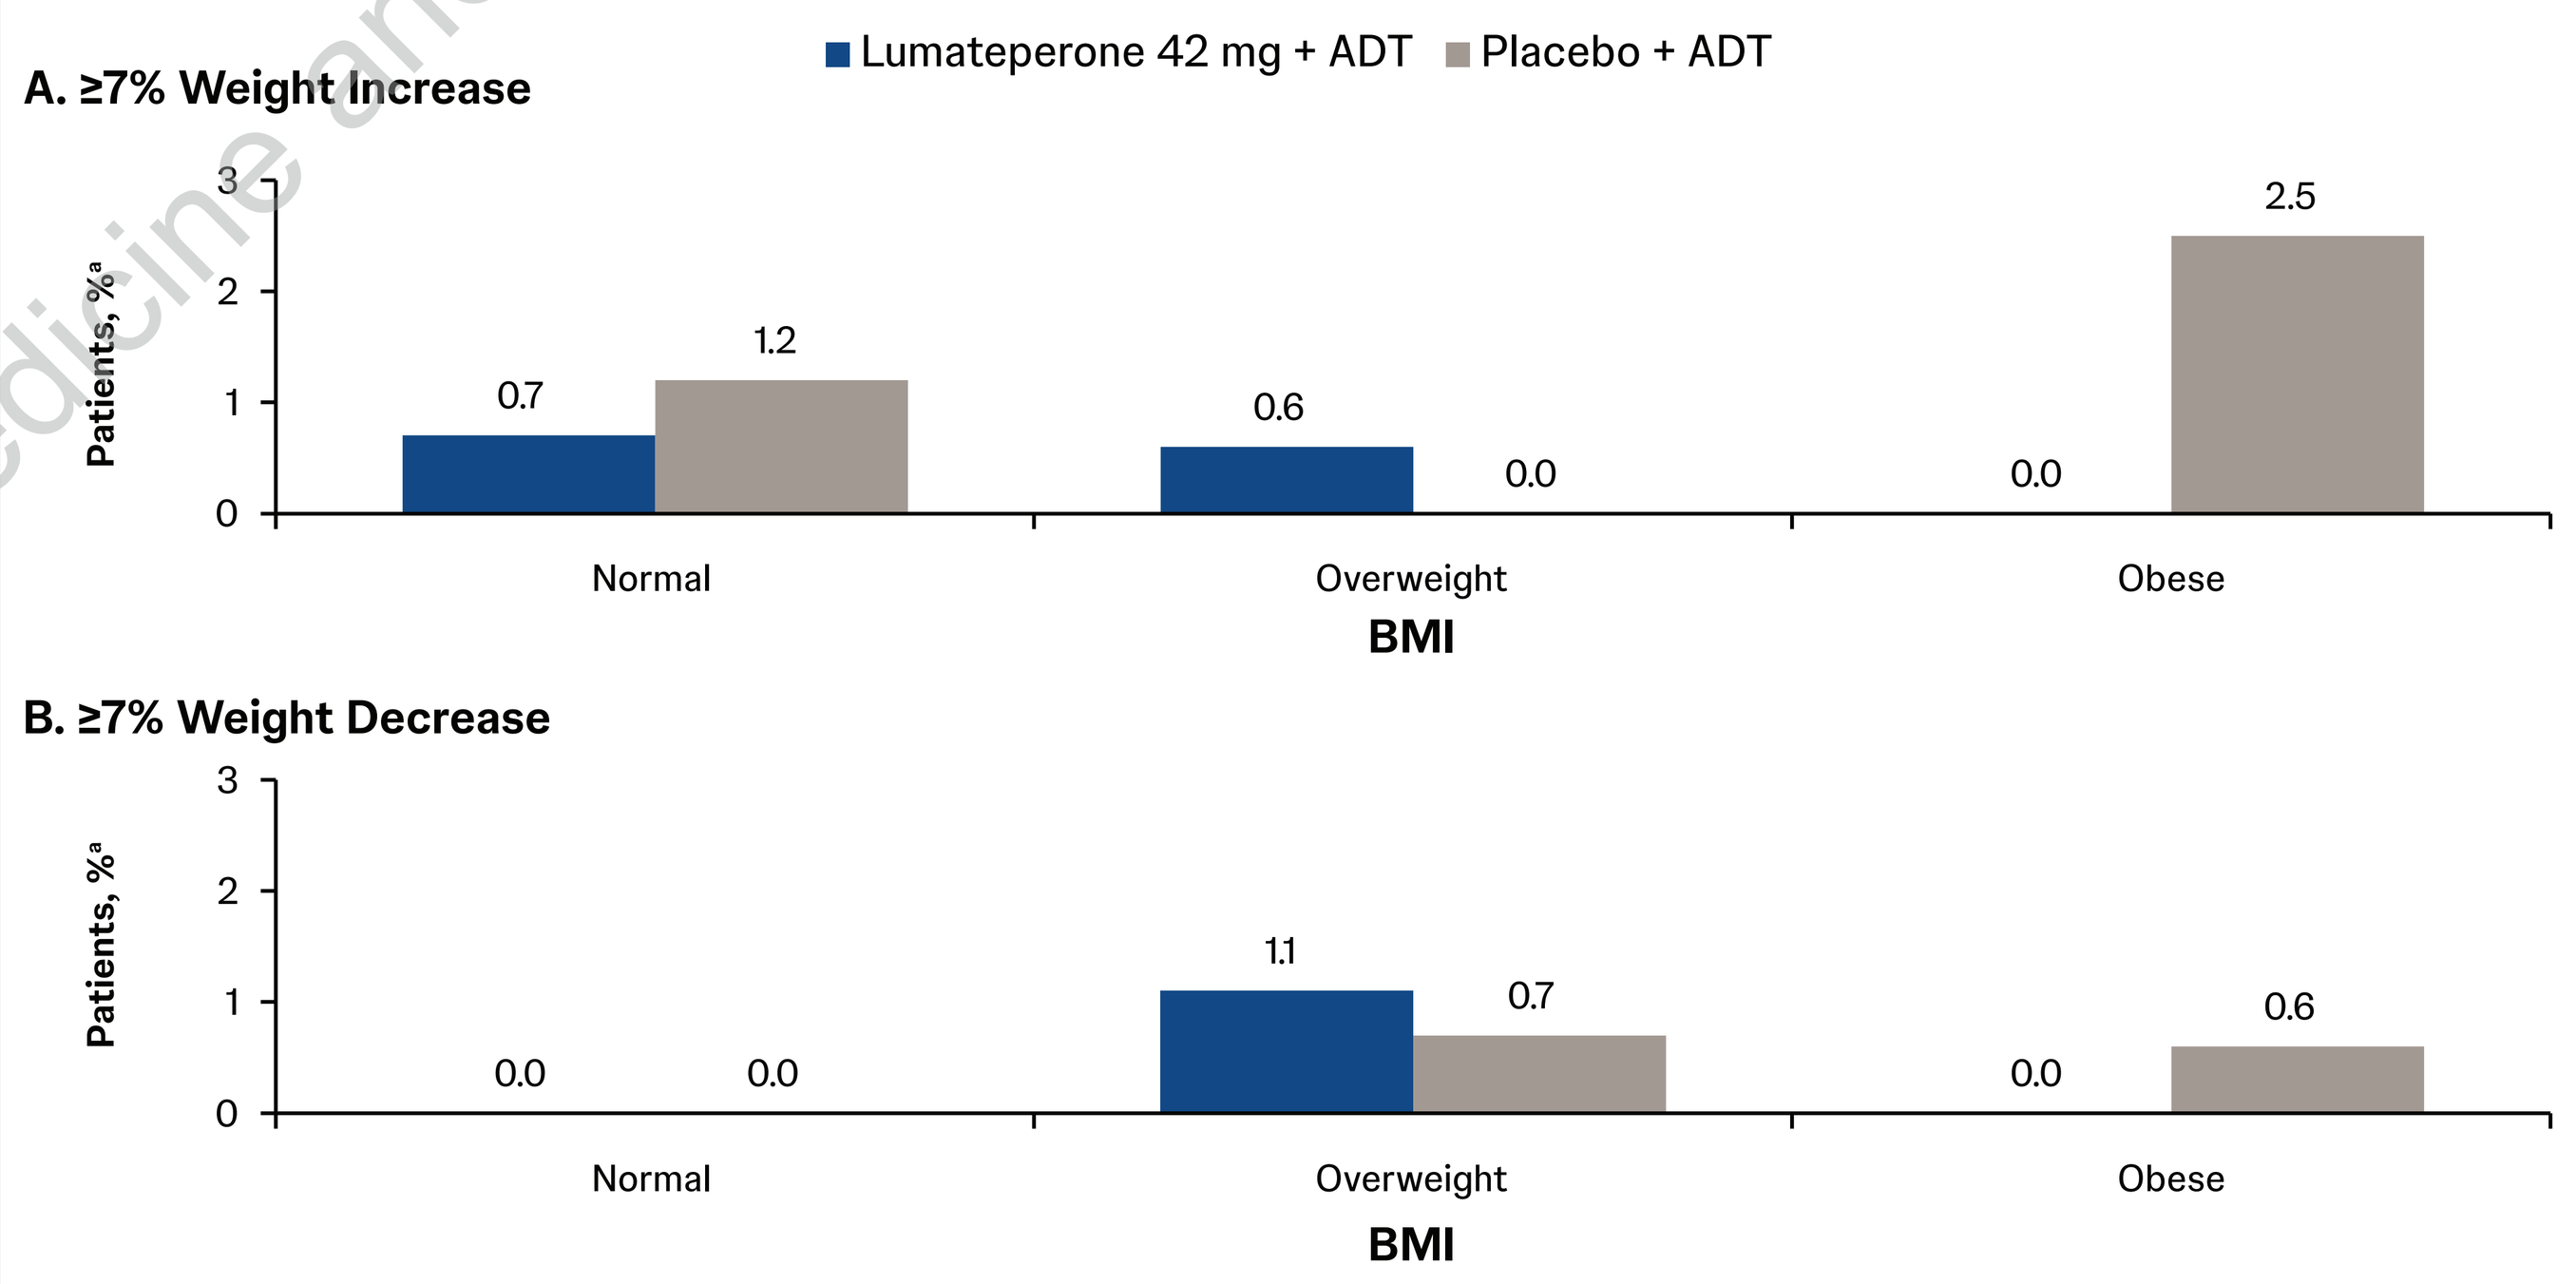
Lumateperone 42 mg + ADT
Placebo + ADT
A. ≥7% Weight Increase
Patients, %a
0.7
1.2
0.6
0.0
0.0
2.5
Normal
Overweight
Obese
BMI
B. ≥7% Weight Decrease
Patients, %a
0.0
0.0
1.1
0.7
0.0
0.6
Normal
Overweight
Obese
BMI

[a]Percentage is n/N1, with n = number of patients who met the criterion at least once during the double-blind treatment period and N1 = number of patients with baseline BMI in the category with ≥1 postbaseline assessment during the double-blind treatment period.
ADT, antidepressant therapy; BMI, body mass index; PCS, potentially clinically significant.

Cardiometabolic Parameters and Prolactin Levels

- Changes in cardiometabolic parameters and prolactin levels at EOT were not clinically relevant across BMI subgroups and treatment arms (**Figure 5**)

Figure 5. Changes in Cardiometabolic Parameters and Prolactin Levels by Baseline BMI Category

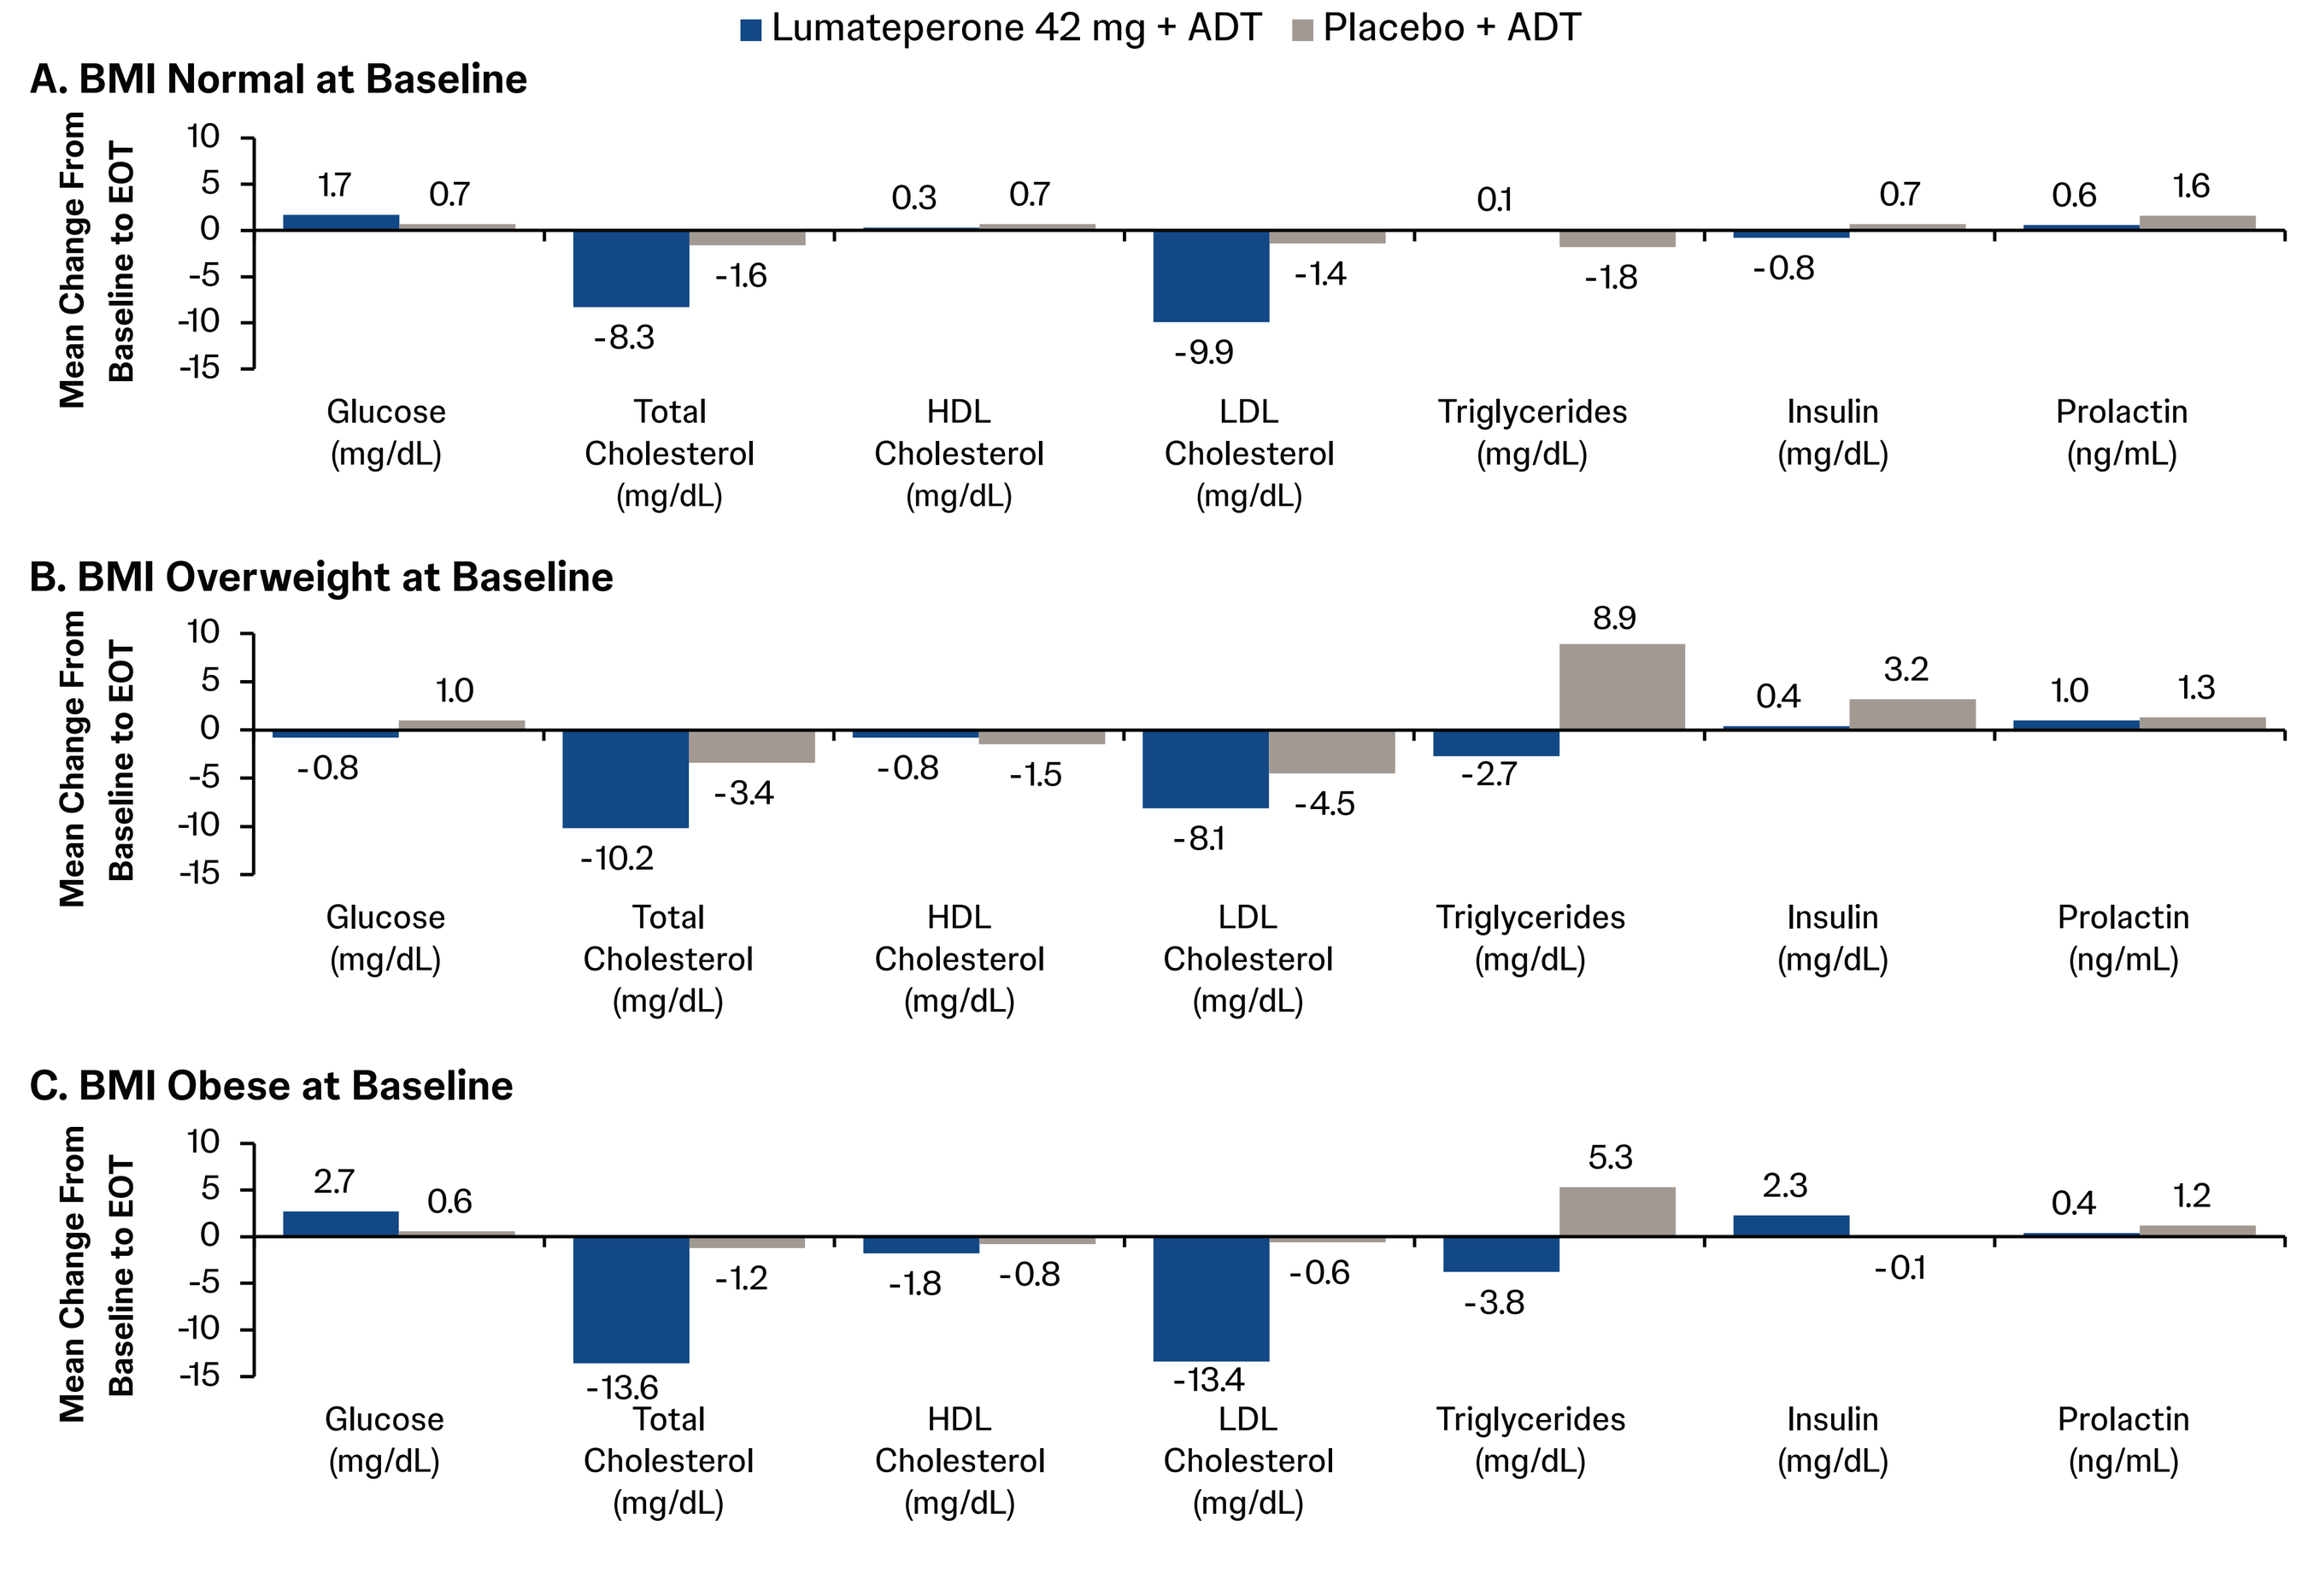
Lumateperone 42 mg + ADT
Placebo + ADT
A. BMI Normal at Baseline
Mean Change From Baseline to EOT
1.7
0.7
-8.3
-1.6
0.3
0.7
-9.9
-1.4
0.1
-1.8
-0.8
0.7
0.6
1.6
Glucose (mg/dL)
Total Cholesterol (mg/dL)
HDL Cholesterol (mg/dL)
LDL Cholesterol (mg/dL)
Triglycerides (mg/dL)
Insulin (mg/dL)
Prolactin (ng/mL)
B. BMI Overweight at Baseline
-0.8
1.0
-10.2
-3.4
-0.8
-1.5
-8.1
-4.5
-2.7
8.9
0.4
3.2
1.0
1.3
C. BMI Obese at Baseline
2.7
0.6
-13.6
-1.2
-1.8
-0.8
-13.4
-0.6
-3.8
5.3
2.3
-0.1
0.4
1.2

ADT, antidepressant therapy; BMI, body mass index; EOT, end of treatment; HDL, high-density lipoprotein; LDL, low-density lipoprotein.

Conclusions

- In this pooled analysis, 6-week treatment with lumateperone 42 mg + ADT had a favorable safety profile
- Minimal changes in body morphology, cardiometabolic parameters, and prolactin levels were observed across BMI subgroups
- Most patients remained in the same BMI category from baseline to EOT
- PCS weight changes were rare with lumateperone 42 mg + ADT
- These results indicate that lumateperone 42 mg may be a well-tolerated adjunctive treatment for patients with MDD with inadequate ADT response

Acknowledgments

The authors thank all study investigators, research staff, and patients for their participation. Medical writing support was provided by Thato Gweja, PhD, of Nucleus Global, an Inizio company, and funded by Intra-Cellular Therapies, a Johnson & Johnson company.

Disclosures

C Chepke has served on the advisory boards of AbbVie, Acadia, Alkermes, Axsome, Biogen, Bristol Myers Squibb, Corium, Eli Lilly, Idorsia, Intra-Cellular Therapies, Jazz, Johnson & Johnson, Lundbeck, Moderna, Neurocrine, Otsuka, Sage, Sumitomo, Teva; advisory boards (spouse): Bristol Myers Squibb, Otsuka; has been a consultant for: AbbVie, Acadia, Alkermes, Axsome, Biogen, Boehringer Ingelheim, Bristol Myers Squibb, Corium, Eli Lilly, Intra-Cellular Therapies, Johnson & Johnson, Lundbeck, Medincell, Moderna, Neurocrine, Otsuka, Sage, Sumitomo, Supernus, Teva; has received research grants from: Acadia, Axsome, Harmony, Neurocrine, Teva; has received speaker/promotional honoraria from: AbbVie, Acadia, Alkermes, Axsome, Bristol Myers Squibb, Corium, Intra-Cellular, Johnson & Johnson, Lundbeck, Luye, Merck, Neurocrine, Otsuka, Sumitomo, Teva; has no stocks/stock options/ownership interest/patents.

WR Earley, R Migliore, and S Madi are full-time employees of Intra-Cellular Therapies, a Johnson & Johnson company. SG Kozauer is a former employee of Intra-Cellular Therapies, a Johnson & Johnson company.

Funding

This study was funded by Intra-Cellular Therapies, a Johnson & Johnson company.

Previously presented at the American Society of Clinical Psychopharmacology (ASCP) Annual Meeting, May 26-29, 2026, Miami, FL, USA.

Neuropsychiatry

Scan the QR code

The QR code is intended to provide scientific information for individual reference, and the information should not be altered or reproduced in any way.

Presented at Psych Congress 2026, September 15-19, 2026, New Orleans, LA, USA

References:

1. Proudman D, et al. *PharmacoEconomics.* 2021;39(6):619-625; 2. Hasin DS, et al. *JAMA Psychiatry.* 2018;75(4):336-346; 3. Baune BT, et al. *Neuropsychiatr Dis Treat.* 2021;17:2995-3006; 4. Arnaud AM, et al. *J Clin Psychiatry.* 2022;83(6):21r14328; 5. Correll CU, et al. *World Psychiatry.* 2017;16:163-180; 6. Vancampfort D, et al. *Psychol Med.* 2014;44:2017-2028; 7. Alva G. *CNS Spectr.* 2023;28(5):521-525; 8. Carvalho AF, et al. *Psychother Psychosom.* 2016;85:270-288; 9. Mago R, et al. *BMC Psychiatry.* 2018;18:1-9; 10. Rafeyan R, et al. *J Clin Psychiatry.* 2020;81(3):27570; 11. Spielmans GI, et al. *PLoS Med.* 2013;10(3):e1001403; 12. Thase ME. *Focus.* 2016;14(2):180-183; 13. Caplyta (lumateperone). Prescribing information. Intra-Cellular Therapies, Inc; 2026; 14. Durgam S, et al. *J Clin Psychiatry.* 2025;86(4):25m15848; 15. Durgam S, et al. *Am J Psychiatry.* 2025;182(12):1072-1082.

Email questions to
Craig Chepke at craig.chepke@gmail.com

This material is distributed for scientific purposes by Johnson & Johnson Innovative Medicine and is not for promotional use.