Lumateperone Treatment of Manic Episodes With or Without Mixed Features in Bipolar I Disorder: Results From a Double-Blind, Placebo-Controlled, Randomized, Phase 3 Trial

Andrew J. Cutler, MD[1]; Willie R. Earley, MD[2]; Robert Hayes, PhD[2]; Changzheng Chen, PhD[2]; Hassan Lakkis, PhD[2]; Maxine Chen, PhD[2]; David Brown, MD[3]; Michael E. Thase, MD[4]

[1]Department of Psychiatry, SUNY Upstate Medical University, Lakewood Ranch, FL, USA; [2]Johnson & Johnson, Titusville, NJ, USA; [3]Community Clinical Research, Inc., Austin, TX, USA; [4]Perelman School of Medicine, University of Pennsylvania, Philadelphia, PA, USA

72

Background

- Bipolar I disorder is characterized by the occurrence of ≥1 manic episode, which may be preceded or followed by hypomanic and/or major depressive episodes (MDE)[1,2]
- More than 90% of patients who experience manic episodes go on to have recurrent mood episodes, and around 60% of manic episodes occur immediately before an MDE[2]
- Bipolar disorder is characterized by high rates and durations of persistent illness, frequent hospitalization, symptom recurrence and rehospitalization, and elevated risk of suicide[3-7]
- Patients with bipolar disorder desire new, effective treatments to stabilize symptoms and reduce suicidal thoughts and with better safety profiles than those currently available[8]
- Lumateperone is an atypical antipsychotic indicated for depressive episodes associated with bipolar I or bipolar II disorder, for schizophrenia, and as adjunctive therapy to ADT in MDD[9]
- Study 451 is the first randomized, double-blind, placebo-controlled trial to evaluate the efficacy and safety of lumateperone 42 mg for the treatment of acute mania in patients with a diagnosis of bipolar I disorder, including those with mixed features as defined in the *Diagnostic and Statistical Manual of Mental Disorders, Fifth Edition, Text Revision* (DSM-5-TR)

Methods

- Study 451 was a 3-week, randomized, double-blind, placebo-controlled trial to investigate the efficacy and safety of lumateperone 42 mg for the treatment of manic episodes with or without mixed features in patients with bipolar I disorder (**Figure 1**; **Table 1**)

Figure 1. Study 451 Design

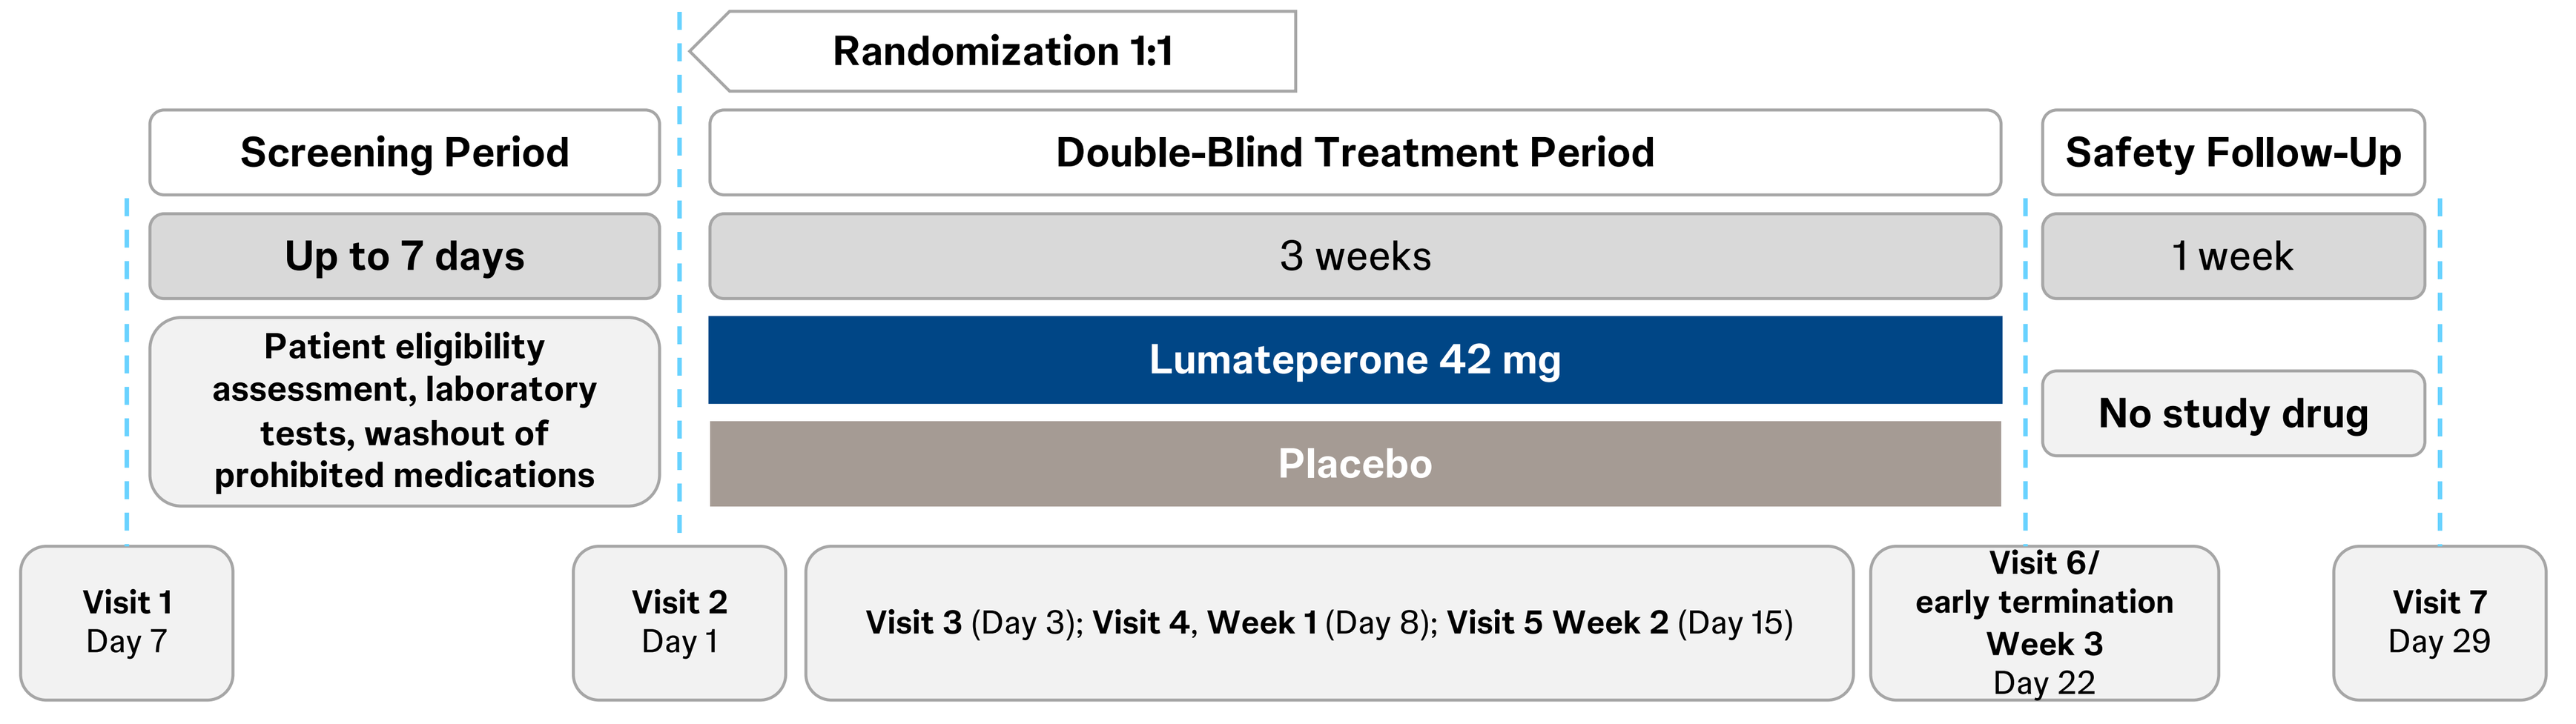
Randomization 1:1
Screening Period
Double-Blind Treatment Period
Safety Follow-Up
Up to 7 days
3 weeks
1 week
Patient eligibility assessment, laboratory tests, washout of prohibited medications
Lumateperone 42 mg
Placebo
No study drug
Visit 1 Day 7
Visit 2 Day 1
Visit 3 (Day 3); Visit 4, Week 1 (Day 8); Visit 5 Week 2 (Day 15)
Visit 6/ early termination Week 3 Day 22
Visit 7 Day 29

Table 1. Study 451 Inclusion and Exclusion Criteria

Inclusion Criteria	• Adults hospitalized because of the current manic episode • DSM-5-TR–diagnosed bipolar I with a current episode of mania or mania with mixed features ≤4 weeks prior to Visit 1 with or without psychotic symptoms • YMRS Total score ≥20 (a Total score ≥25 was considered severely ill[10]) plus • YMRS score of ≥4 on two key items at Visits 1 and 2: irritability, speech, content, or disruptive/aggressive behavior plus • CGI-S score ≥4 at Visits 1 and 2
Exclusion Criteria	• Patient's primary DSM-5 psychiatric diagnosis was anything other than bipolar disorder • They were experiencing their first manic episode • They had a significant risk for suicidal behavior in the opinion of the investigator

CGI-S, Clinical Global Impression–Severity; DSM-5, *Diagnostic and Statistical Manual of Mental Disorders, Fifth Edition*; DSM-5-TR, *Diagnostic and Statistical Manual of Mental Disorders, Fifth Edition, Text Revision;* YMRS, Young Mania Rating Scale.

- Efficacy assessments
 - The primary endpoint was the change from baseline (ie, Day 1, prerandomization) to end of Week 3 (Day 22) in YMRS Total score
 - Clinically meaningful effects (additional efficacy measures) were assessed as YMRS response (≥50% decrease from baseline in YMRS Total score) and remission (YMRS Total score ≤12)
 - The key secondary endpoint was the change from baseline to end of Week 3 (Day 22) in CGI-S score
 - Endpoint results were subjected to fixed sequence testing: Primary → key secondary → Week 1 (Day 8) YMRS → Day 3 YMRS
- Safety assessment
 - Safety and tolerability were evaluated throughout the treatment period and for 1 week afterward
- Study populations
 - Safety population: all randomized patients who received ≥1 dose of study treatment
 - Intent-to-treat (ITT) population includes all randomized patients who received ≥1 dose of study drug and had a baseline YMRS Total score

Results

Patient population

- Of 354 patients treated (lumateperone, n=177; placebo, n=177), 327 (92.4%) completed treatment (safety and ITT populations)
- The mean age at the start of the study was 46.6 years, and 55.6% were female (**Table 2**)
- More than half of patients had experienced ≤5 lifetime manic episodes (58.8%), and 15.3% were diagnosed with mixed features according to the DSM-5 (**Table 2**)
- Mean YMRS Total score at baseline indicated severe mania in both groups (33.2 in the lumateperone 42-mg group and 32.1 in the placebo group) (**Table 2**)
- Baseline demographics and psychiatric history were balanced between treatment groups

Table 2. Baseline Demographics and Clinical Characteristics (Safety Population)

	Lumateperone 42 mg (n=177)	Placebo (n=177)	Total (N=354)
Age, mean (SD), years	47.2 (13.2)	45.9 (11.5)	46.6 (12.4)
Female, n (%)	95 (53.7)	102 (57.6)	197 (55.6)
Race, n (%)			
White	115 (65.0)	120 (67.8)	235 (66.4)
Black or African American	41 (23.2)	37 (20.9)	78 (22.0)
Asian	15 (8.5)	15 (8.5)	30 (8.5)
Other	1 (0.6)	1 (0.6)	2 (0.6)
Ethnicity, n (%)			
Hispanic or Latino	36 (20.3)	36 (20.3)	72 (20.3)
Not Hispanic or Latino	141 (79.7)	141 (79.7)	282 (79.7)
Any specifier associated with current episode, n (%)			
Yes	75 (42.4)	73 (41.2)	148 (41.8)
Mixed features specifier[a]			
Yes	24 (13.6)	30 (16.9)	54 (15.3)
No	51 (28.8)	43 (24.3)	94 (26.6)
Psychotic features specifier[a]			
Yes	63 (35.6)	51 (28.8)	114 (32.2)
No	12 (6.8)	22 (12.4)	34 (9.6)
No	102 (57.6)	104 (58.8)	206 (58.2)
Time since start of current manic episode, mean (SD), days	15.9 (5.29)	16.2 (5.83)	16.0 (5.56)
Lifetime manic episodes, n (%)			
≤5	102 (57.6)	106 (59.9)	208 (58.8)
>5 and ≤10	30 (16.9)	39 (22.0)	69 (19.5)
>10 and ≤20	25 (14.1)	19 (10.7)	44 (12.4)
>20	14 (7.9)	8 (4.5)	22 (6.2)
Unknown	6 (3.4)	5 (2.8)	11 (3.1)
Age at first bipolar disorder diagnosis, mean (SD), years	29.3 (11.4)	28.5 (9.4)	28.9 (10.4)
YMRS Total score, mean (SD)	33.2 (5.5)	32.1 (5.6)	–
CGI-S, mean (SD)	4.9 (0.6)	4.8 (0.6)	–

[a]Percentages are based on the total number of patients in the respective treatment arm or the total population.
CGI-S, Clinical Global Impression–Severity; YMRS, Young Mania Rating Scale.

Efficacy

- Lumateperone 42 mg demonstrated statistical superiority to placebo for the change in YMRS Total score from baseline at Day 3 (*P*<.001) (**Figure 2**)
- The superiority of lumateperone 42 mg over placebo was maintained through to the end of Week 3 (Day 22) with a least squares mean difference (LSMD) of −4.8 (effect size [ES], −0.69; *P*<.0001) (**Figure 2**)

Figure 2. Change From Baseline in YMRS Total Score (ITT Population)

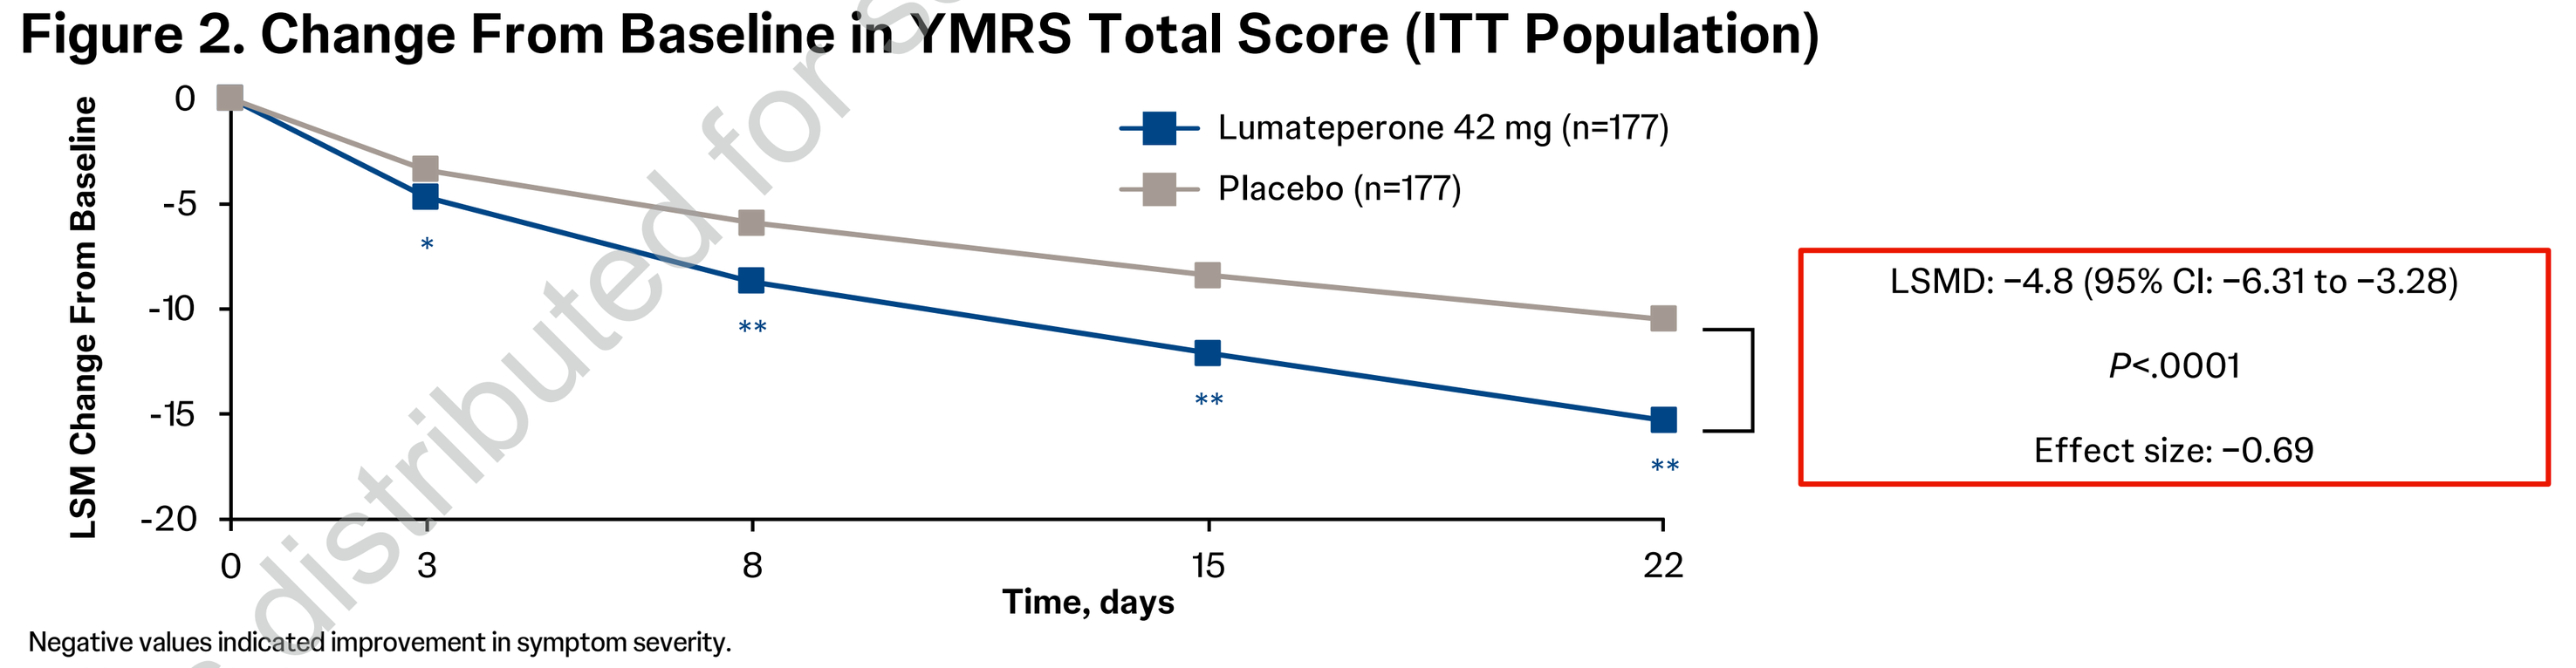
LSM Change From Baseline
0
-5
-10
-15
-20
0
3
8
15
22
Time, days
Lumateperone 42 mg (n=177)
Placebo (n=177)
*
**
LSMD: −4.8 (95% CI: −6.31 to −3.28)
P<.0001
Effect size: −0.69

Negative values indicated improvement in symptom severity.
*P<.001 **P<.0001. LSMD. Analysis of covariance in the ITT population.
CI, confidence interval; ITT, intent-to-treat; LSM, least squares mean; LSMD, least squares mean difference; YMRS, Young Mania Rating Scale.

- Lumateperone 42 mg vs placebo showed significantly greater response rates (YMRS Total score ≥50% decrease; 45.8% vs 20.9%; *P*<.0001; **Figure 3**)
- Lumateperone 42 mg vs placebo showed significantly greater remission rates (YMRS Total score ≤12; 30.5% vs 16.4%; *P*=.0009; **Figure 3**)
- The number needed to treat for lumateperone 42 mg was 4.0 to achieve response and 7.1 to achieve remission

Figure 3. YMRS Total Score Response and Remission Rates at Day 22 (ITT Population)

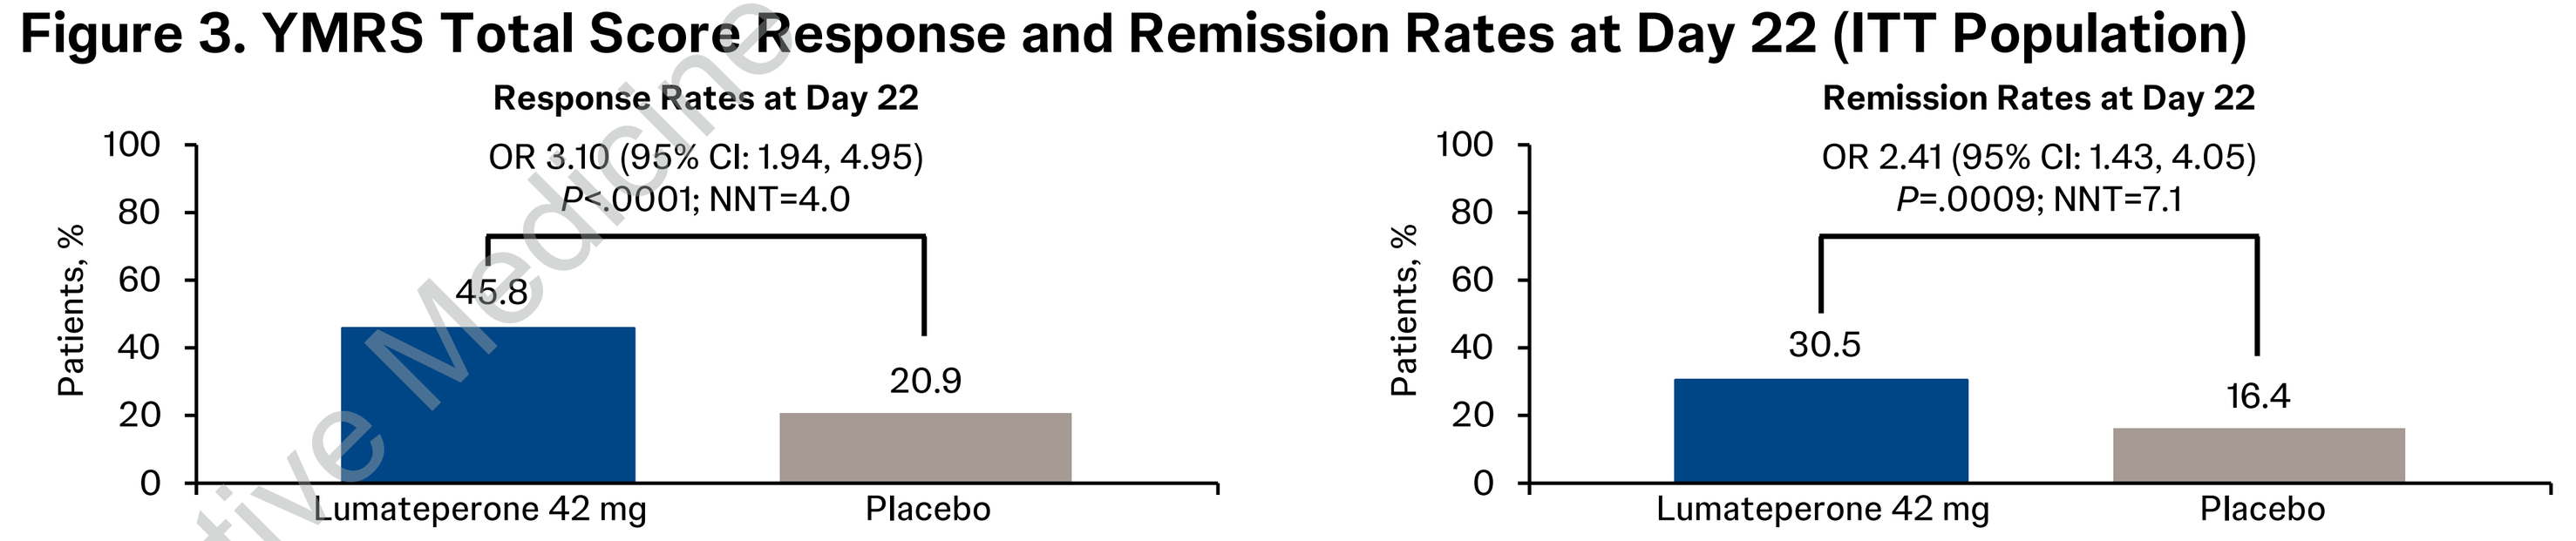
Response Rates at Day 22
OR 3.10 (95% CI: 1.94, 4.95)
P<.0001; NNT=4.0
Patients, %
100
80
60
40
20
0
45.8
20.9
Lumateperone 42 mg
Placebo
Remission Rates at Day 22
OR 2.41 (95% CI: 1.43, 4.05)
P=.0009; NNT=7.1
30.5
16.4

Response was defined as ≥50% decrease from baseline YMRS Total score at Day 22; remission was defined as a YMRS Total score ≤12 at Day 22.
CI, confidence interval; ITT, intent-to-treat; NNT, number needed to treat; OR, odds ratio: YMRS, Young Mania Rating Scale.

- Lumateperone 42 mg demonstrated statistical superiority to placebo for the change in CGI-S score from baseline at Day 8 (*P*<.001) (**Figure 4**)
- The superiority of lumateperone 42 mg over placebo was maintained through to the end of Week 3 (Day 22) with a LSMD of −0.5 (ES, −0.62; *P*<.0001) (**Figure 4**)

Figure 4. Change From Baseline in CGI-S Score (ITT Population)

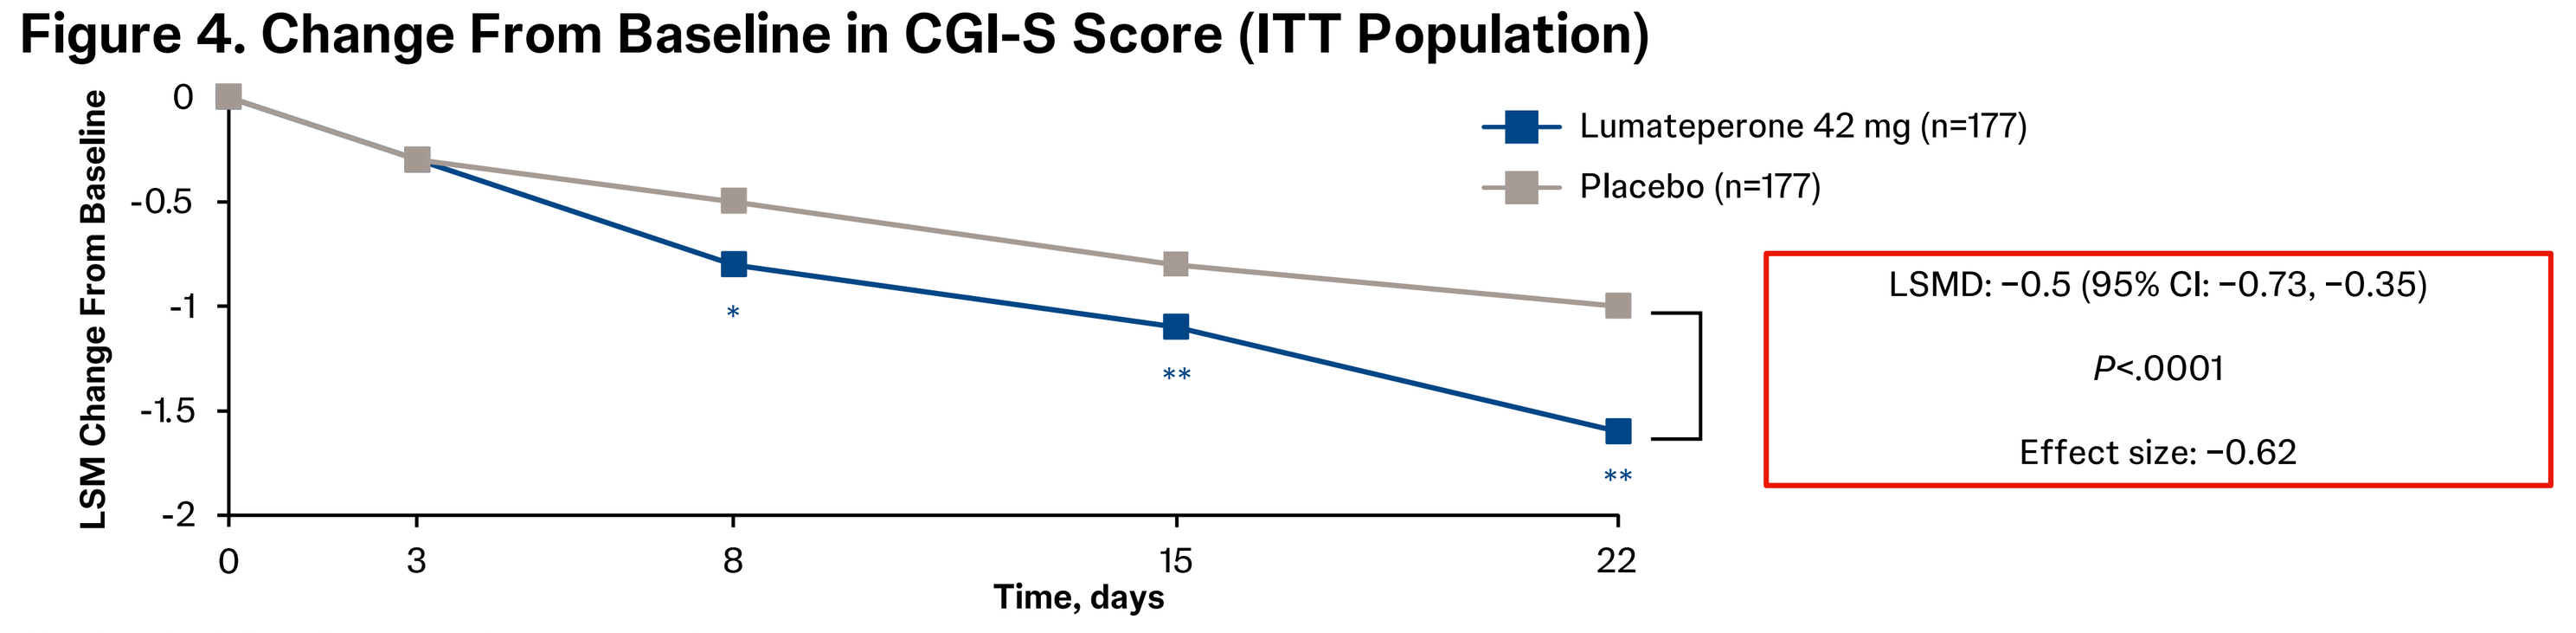
LSM Change From Baseline
0
-0.5
-1
-1.5
-2
0
3
8
15
22
Time, days
Lumateperone 42 mg (n=177)
Placebo (n=177)
*
**
LSMD: −0.5 (95% CI: −0.73, −0.35)
P<.0001
Effect size: −0.62

Negative values indicated improvement in symptom severity.
*P<.001 **P<.0001. LSMD. Analysis of covariance in the ITT population.
CI, confidence interval ; CGI-S, Clinical Global Impression–Severity; ITT, intent-to-treat; LSM, least squares mean; LSMD, least squares mean difference.

Safety

- The safety profile of lumateperone 42 mg did not reveal any unexpected findings
- There were 2 serious adverse events (SAEs) and 1 death in the lumateperone 42 mg group
 - The cause of death was considered by the investigator as likely multifactorial and not directly due to treatment with lumateperone 42 mg; the same patient also experienced an SAE of rhabdomyolysis, which resolved prior to death
 - The other SAE was an occurrence of bipolar disorder
- Two AEs were found to occur in ≥5% of patients receiving lumateperone 42 mg and were ≥2× those in the placebo group: dry mouth (7.9% vs 3.4%) and nausea (7.9% vs 2.3%) (**Table 3**)
- The incidence of somnolence (4.0%) was lower than that seen in studies of lumateperone in patients with MDD or bipolar depression (12.0% to 15.7%)[11,12]
- Discontinuations because of AEs were in <2% in the lumateperone 42 mg group and <3% in the placebo group

Table 3. TEAEs With Frequency ≥2% and Greater Rate Than Placebo Throughout the Double-Blind Treatment Phase

Preferred Term, n (%)	Lumateperone 42 mg (n=177)	Placebo (n=177)
Headache	27 (15.3)	14 (7.9)
Dry mouth[a]	14 (7.9)	6 (3.4)
Nausea[a]	14 (7.9)	4 (2.3)
Dizziness	8 (4.5)	5 (2.8)
Somnolence	7 (4.0)	2 (1.1)
Anxiety	6 (3.4)	5 (2.8)
Diarrhea	6 (3.4)	1 (0.6)
Agitation	5 (2.8)	1 (0.6)
Vomiting	4 (2.3)	3 (1.7)
Hyperinsulinemia	4 (2.3)	2 (1.1)
Nasopharyngitis	4 (2.3)	0
Vulvovaginal mycotic infection[b]	2 (2.1)	0

[a]≥5% in the lumateperone 42 mg group and at least 2× placebo.
[b]Female patient–specific adverse event; % based on number of female patients.
TEAE, treatment-emergent adverse event.

Conclusions

- Lumateperone 42 mg vs placebo substantially and rapidly improved acute manic symptoms in patients with bipolar I, with and without mixed features
- Effects on YMRS Total score were significant from Day 3 and were maintained throughout the treatment period
- More patients receiving lumateperone 42 mg achieved YMRS response and remission by the end of treatment than those receiving placebo
- Improvements in CGI-S scores were significantly greater in patients receiving lumateperone 42 mg vs placebo at Day 8 and were maintained to the end of treatment
- Lumateperone 42 mg had a consistent safety and tolerability profile and was well tolerated, with no new safety findings and low rates of discontinuation

Acknowledgments

The authors thank all study investigators, research staff, and patients for their participation. Medical writing support was provided by Apurva H. Davé, PhD, of Nucleus Global, an Inizio company, funded by Intra-Cellular Therapies, a Johnson & Johnson company.

Disclosures

This study was sponsored by Johnson & Johnson.

AJ Cutler has served as a consultant/on an advisory board for AbbVie, Acadia, Actinogen, Alfasigma, Alkermes, Anavex Life Sciences, Arrivo BioVentures, Autobahn Therapeutics, Axsome, Aytu BioPharma, Bristol Myers Squibb, Collegium Pharmaceutical, Definium Therapeutics, 4M Therapeutics, Helus Pharma, Incannex Healthcare, Intra-Cellular Therapies, J&J Innovative Medicine, Jazz Pharma, Knight Therapeutics, Kye Pharmaceuticals, Kuvatris Therapeutics, LivaNova, Lundbeck, Luye Pharma, MapLight Therapeutics, Mentavi, Neumora, Neurocrine Biosciences, Noven, Otsuka, Sensorium Therapeutics, Sirtsei Pharmaceuticals, Supernus, Teva, Thynk, Transneural Therapeutics, Tris Pharma, Vanda Pharmaceuticals, and Vistagen; served on a speaker's bureau for AbbVie, Alfasigma, Alkermes, Axsome, Aytu BioPharma, Bristol Myers Squibb, Collegium Pharmaceutical, Intra-Cellular Therapies, J&J Innovative Medicine, Knight Therapeutics, Kye Pharmaceuticals, Lundbeck, Luye Pharma, Neurocrine Biosciences, Noven, Otsuka, Supernus, Teva, Tris Pharma, and Vanda Pharmaceuticals; served on a data safety monitoring board for COMPASS Pathways; and has stock options/equity in EMA Wellness, Evolution Research Group, 4M Therapeutics, Sensorium Therapeutics, Transneural Therapeutics.

WR Earley, R Hayes, C Chen, H Lakkis, and M Chen are employees of Johnson & Johnson.

D Brown has conducted clinical trials for AbbVie, Boehringer Ingelheim, Bristol Myers Squibb, Draig Therapeutics, Johnson & Johnson, Neurocrine Biosciences, PleoPharma, Rapport Therapeutics, Teva, and Vanda Pharmaceuticals and is a speaker for Bristol Myers Squibb.

ME Thase served as an advisor or a consultant for Autobahn Therapeutics; Axsome Therapeutics, Inc.; Clexio Biosciences; Gerson Lehrman; GH Therapeutics; H. Lundbeck, A/S; Janssen Pharmaceuticals, Inc.; Johnson & Johnson; Luye Pharma Group, Ltd.; Merck & Company, Inc.; Object Pharma; Otsuka Pharmaceutical Company, Ltd.; Atai-Beckley; Pfizer, Inc.; Sage Pharmaceuticals; Seelos Therapeutics; Takeda Pharmaceutical Company, Ltd.; has received grants from Acadia Inc.; Alkermes; Axsome Therapeutics Inc.; Intra-Cellular Therapies; Janssen Pharmaceuticals, Inc.; Myriad; National Institute of Mental Health; Otsuka Pharmaceutical Company, Ltd.; Patient-Centered Outcomes Research Institute (PCORI); Takeda Pharmaceutical Company, Ltd.; and has received royalties from the American Psychiatric Association Foundation; Guilford Publications; Herald House; Wolters Kluwer; and W.W. Norton & Company, Inc.

Neuropsychiatry

Scan the QR code
The QR code is intended to provide scientific information for individual reference, and the information should not be altered or reproduced in any way.

Presented at Psych Congress 2026, September 15–19, 2026, New Orleans, LA, USA

References:
1. American Psychiatric Association. DSM-5. 2013; 2. American Psychiatric Association. DSM-5 Text Revision. 2022; 3. Forte A, et al. *J Affect Disord*. 2015;178:71-78; 4. Ng-Mak D, et al. *Curr Med Res Opin*. 2019:35:211-219; 5. Vázquez GH, et al. *Eur Neuropsychopharmacol*. 2015;10:1501-1512; 6. Bozzay ML, et al. *Psychiatry Res*. 2019;281:112559; 7. Dome P, et al. *Medicina.* 2019;55:403; 8. Busch C, et al. *J Affective Disord Rep*. 2026;25:101114; 9. Caplyta (lumateperone). Prescribing information. Intra-Cellular Therapies, Inc; 2026; 10. Lukaiewicz M, et al. *Int J Methods Psychiatr Res*. 2013;22:46-58; 11. Durgam S, et al. *J Clin Psychopharmacol.* 2025;45:67-75; 12. Durgam S, et al. *CNS Spectrums*. 2025;30(e80):1-7.

Email questions to Andrew J. Cutler at acutler@ajcmd.com

This material is distributed for scientific purposes by Johnson & Johnson Innovative Medicine and is not for promotional use.