113

Lumateperone 42 mg in Major Depressive Disorder: Demographic and Clinical Subgroups Efficacy Analysis in a Phase 3 Randomized Placebo-Controlled Trial

Suresh Durgam, MD[1]; Raffaele Migliore, MA[2]; Willie R. Earley, MD[2]; Yifan Mo, PhD[2]; Hassan Lakkis, PhD[2]; Susan G. Kornstein, MD[3]

[1]Former employee of Intra-Cellular Therapies, a Johnson & Johnson company, Bedminster, NJ, USA; [2]Intra-Cellular Therapies, a Johnson & Johnson company, Bedminster, NJ, USA; [3]Department of Psychiatry, Virginia Commonwealth University School of Medicine, Richmond, VA, USA

Background

- Major depressive disorder (MDD) is a highly disabling, complex, and heterogenous disease that burdens a diverse patient population[1-3]
- In patients with MDD with inadequate response to antidepressant therapy (ADT), adjunctive atypical antipsychotic treatment is a recognized treatment strategy, warranting the need to evaluate its use across demographic and disease characteristics[4]
- Lumateperone is an atypical antipsychotic indicated for the treatment of MDD in adults as adjunct to ADT; the treatment of schizophrenia in adults; and the treatment of depressive episodes associated with bipolar I or II disorder (bipolar depression) in adults, as monotherapy and as adjunctive therapy with lithium or valproate[5]
 - Lumateperone is a potent serotonin 5-HT_{2A} receptor antagonist, a dopamine D_2 receptor presynaptic partial agonist and postsynaptic antagonist, a D_1 receptor-dependent indirect modulator of glutamatergic AMPA and NMDA currents, and a serotonin reuptake inhibitor[6]
 - This novel mechanism of action with multimodal effects may confer robust efficacy with improved tolerability compared with current treatment options
- The efficacy and safety of lumateperone 42 mg adjunctive to ADT were established in two Phase 3 randomized, double-blind, placebo-controlled trials (Study 501 [NCT04985942] and Study 502 [NCT05061706]) in patients with MDD with inadequate ADT response[7,8]
 - In Study 501, patients who received lumateperone 42 mg + ADT demonstrated consistent and clinically meaningful improvements in depression symptoms and disease severity compared with placebo + ADT across subgroups[9]
- In this post hoc analysis of Study 502, the efficacy of lumateperone 42 mg + ADT vs placebo + ADT was evaluated within various demographic and clinical subgroups of patients with MDD with inadequate ADT response

Methods

- Eligible patients (aged 18-65 years) met DSM-5 criteria for MDD with inadequate response to 1-2 ADT courses in the current major depressive episode and had Montgomery-Åsberg Depression Rating Scale (MADRS) Total score ≥24, Clinical Global Impression–Severity (CGI-S) score ≥4, and Quick Inventory of Depressive Symptomatology-Self Report-16 item (QIDS-SR-16) Total score ≥14
 - Inadequate response to ADT was defined as <50% improvement with ≥6 weeks ADT monotherapy, as confirmed by the Antidepressant Treatment Response Questionnaire
- Patients were randomized 1:1 to 6-week oral lumateperone 42 mg or placebo adjunctive to ADT
- Efficacy was assessed in the overall population for the primary endpoint (change in MADRS Total score from baseline to Day 43) and key secondary endpoint (change in CGI-S score from baseline to Day 43) using a mixed-effects model for repeated measures (MMRM)
 - The subgroup analysis of these endpoints was performed by demographic and disease characteristic subgroups using an MMRM (**Table 1**)
- Change in QIDS-SR-16 Total score from baseline to Day 43 was also assessed in the overall population and subgroups using an analysis of covariance

Table 1. Demographic and Baseline Disease Characteristic Subgroups

Demographic subgroups	Baseline disease characteristic subgroups
• Age: ≤40 or >40 years • Sex: male or female • Race: White or non-White • Ethnicity: Hispanic/Latino or not Hispanic/Latino • Region: US or non-US	• Disease severity: MADRS Total score <32 or ≥32 • Type of ADT: SSRI or SNRI/other • Number of ADT failures in the current episode: 1 or 2 • Presence/absence of DSM-5–defined anxious distress

ADT, antidepressant therapy; DSM-5, *Diagnostic and Statistical Manual of Mental Disorders, Fifth Edition*; MADRS, Montgomery-Åsberg Depression Rating Scale; SNRI, serontonin-norepinephrine reuptake inhibitor; SSRI, selective serotonin reuptake inhibitor.

Results

Patient Population

- The modified intent-to-treat (mITT) population comprised 469 patients (lumateperone + ADT, n=232; placebo + ADT, n=237)
- Baseline demographics and clinical characteristics were similar between treatment groups
 - The mean age of patients was 46 years
 - The majority of patients were female (69.6%) and White (95.4%)

Efficacy

- The primary and key secondary endpoints were met, with lumateperone + ADT significantly improving MADRS Total score (least squares mean difference vs placebo [LSMD], −4.5; effect size [ES], −0.56; *P*<.0001) and CGI-S score (LSMD, −0.5; ES, −0.51; *P*<.0001), respectively, at Day 43 vs placebo + ADT in the mITT population
- Lumateperone + ADT also significantly improved QIDS-SR-16 Total score at Day 43 in the intent-to-treat (ITT) population (LSMD, −2.2; ES, −0.45; *P*<.0001)

Subgroup Analysis

- Lumateperone + ADT significantly improved MADRS Total score from baseline to Day 43 compared with placebo + ADT across all demographic subgroups assessed except for race (White, *P*<.0001; non-White, *P*=.8717) and ethnicity (Hispanic/Latino, *P*=.1468; not Hispanic/Latino, *P*<.0001) (**Figure 1A**)
- Significant improvements in MADRS Total score from baseline to Day 43 were seen with lumateperone + ADT vs placebo + ADT across all baseline disease characteristics evaluated (**Figure 1B**)
- Significant improvement in CGI-S scores from baseline to Day 43 was consistently demonstrated with lumateperone + ADT vs placebo + ADT across all demographic and clinical subgroups, except for race (White, *P*<.0001; non-White, *P*=.8376) and ethnicity (Hispanic/Latino, *P*=.1884; not Hispanic/Latino, *P*<.0001) (**Figures 2A** and **2B**)
- Lumateperone + ADT significantly improved QIDS-SR-16 Total scores from baseline to Day 43 in the age and sex demographic subgroups and across all baseline disease characteristic subgroups compared with placebo + ADT (**Figures 3A** and **3B**)
 - Significance was not reached for race (White, *P*<.0001; non-White, *P*=.6556), ethnicity (Hispanic or Latino, *P*=.2585; not Hispanic or Latino, *P*<.0001), or region (US, *P*=.4518; non-US, *P*<.0001)

Figure 1. Change From Baseline to Day 43 in MADRS Total Score Among (A) Demographic and (B) Disease Characteristic Subgroups (mITT Population)

A. Demographic Subgroups

	Lumateperone 42 mg + ADT n/N1	Placebo + ADT n/N1	LSMD (95% CI)	Effect Size
≤40 years	64/77	66/74	−4.3 (−7.05, −1.62)	−0.54
>40 years	139/155	155/163	−4.6 (−6.45, −2.81)	−0.58
Male	58/70	66/72	−4.0 (−6.75, −1.23)	−0.50
Female	145/162	155/165	−4.8 (−6.56, −2.97)	−0.59
White	198/226	208/222	−4.5 (−6.07, −3.00)	−0.57
Non-White	5/6	13/15	−0.7 (−8.90, 7.55)	−0.08
Hispanic/Latino	32/36	30/33	−2.9 (−6.88, 1.03)	−0.36
Not Hispanic/Latino	171/196	191/204	−4.8 (−6.39, −3.13)	−0.59
US	57/75	65/75	−3.5 (−6.32, −0.64)	−0.42
Non-US	146/157	156/162	−4.8 (−6.70, −2.98)	−0.58

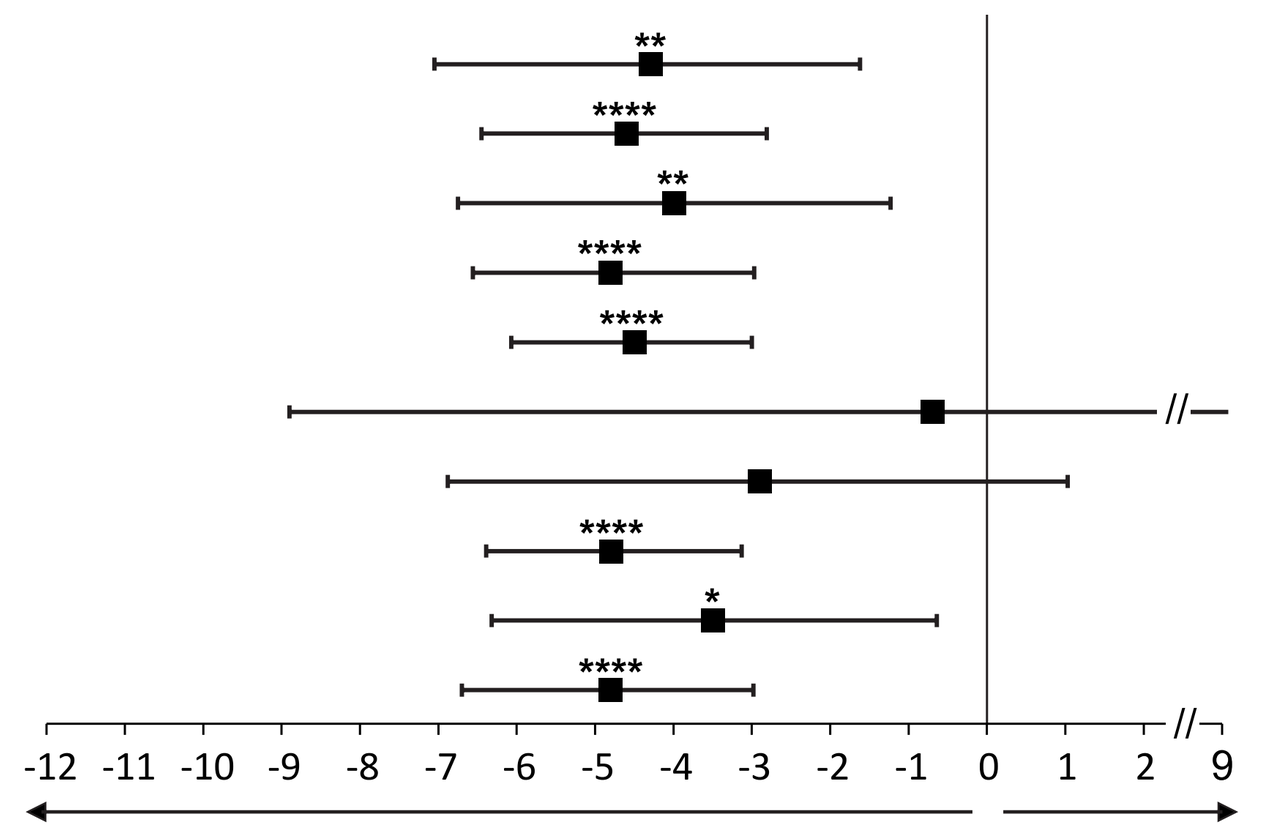
**

**

*

-12 -11 -10 -9 -8 -7 -6 -5 -4 -3 -2 -1 0 1 2 9
Favors lumateperone + ADT
LSMD (95% CI) vs placebo
Favors placebo + ADT

B. Disease Characteristic Subgroups

	Lumateperone 42 mg + ADT n/N1	Placebo + ADT n/N1	LSMD (95% CI)	Effect Size
Baseline MADRS Total score <32	121/137	118/124	−4.6 (−6.64, −2.62)	−0.58
Baseline MADRS Total score ≥32	82/95	103/113	−4.4 (−6.70, −2.12)	−0.55
SSRI	133/153	131/143	−4.3 (−6.24, −2.45)	−0.54
SNRI or other	70/79	90/94	−4.6 (−7.14, −2.12)	−0.58
1 ADT failure	153/175	166/178	−3.3 (−5.01, −1.57)	−0.41
2 ADT failures	50/57	55/59	−8.4 (−11.37, −5.33)	−1.05
Anxious distress at baseline	79/88	82/91	−4.7 (−7.19, −2.28)	−0.59
No anxious distress at baseline	124/144	139/146	−4.4 (−6.30, −2.47)	−0.55

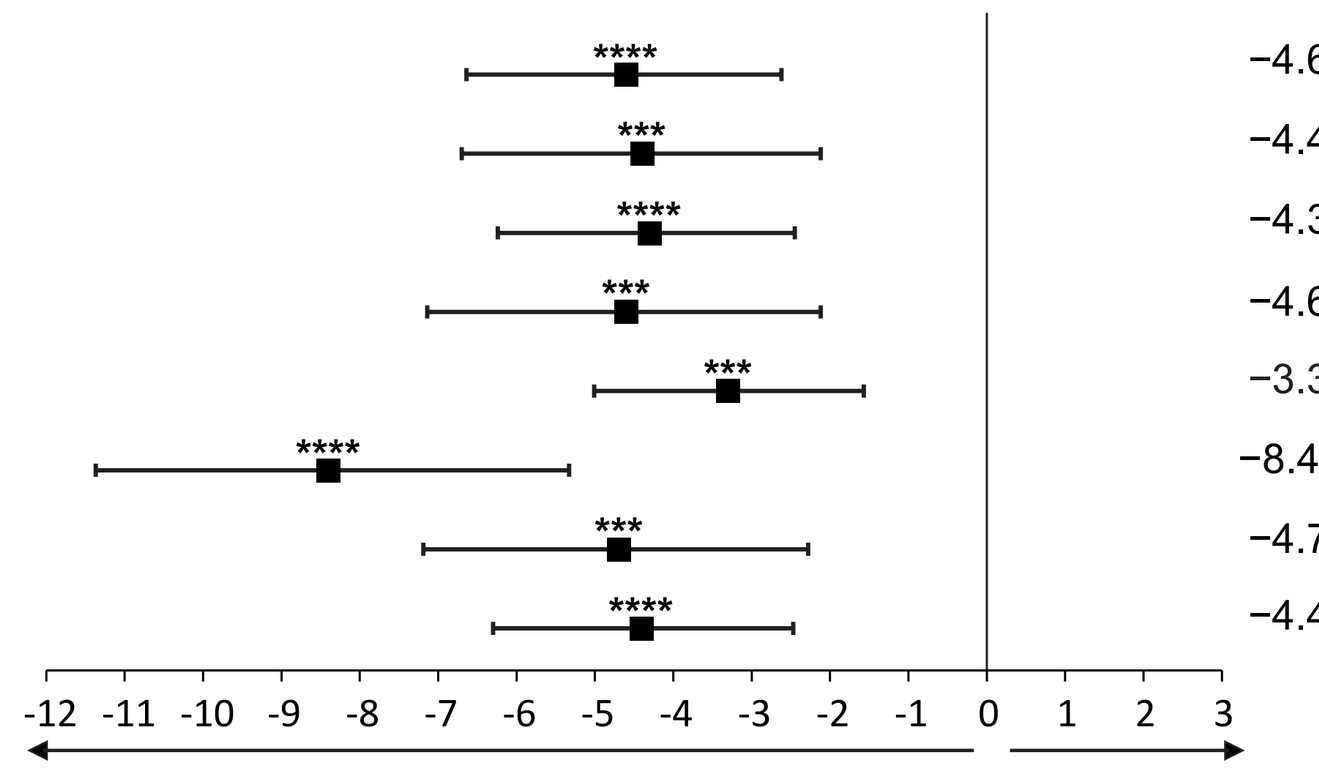
-12 -11 -10 -9 -8 -7 -6 -5 -4 -3 -2 -1 0 1 2 3
Favors lumateperone + ADT
LSMD (95% CI) vs placebo
Favors placebo + ADT

P*<.05 *P*<.01 ****P*<.001 *****P*<.0001. MMRM in the mITT population.
n=Number of patients with Day 43 assessment; N1=Number of patients in each group at baseline.
ADT, antidepressant therapy; LSMD, least squares mean difference; MADRS, Montgomery-Åsberg Depression Rating Scale; mITT, modified intent-to-treat; MMRM, mixed-effects model for repeated measures; SNRI, serotonin-norepinephrine reuptake inhibitor; SSRI, selective serotonin reuptake inhibitor.

Figure 2. Change From Baseline to Day 43 in CGI-S Score Among (A) Demographic and (B) Disease Characteristic Subgroups (mITT Population)

A. Demographic Subgroups

	Lumateperone 42 mg + ADT n/N1	Placebo + ADT n/N1	LSMD (95% CI)	Effect Size
≤40 years	64/77	66/74	−0.4 (−0.79, −0.10)	−0.43
>40 years	139/155	155/163	−0.6 (−0.80, −0.33)	−0.55
Male	58/70	66/72	−0.5 (−0.83, −0.12)	−0.46
Female	145/162	155/165	−0.5 (−0.78, −0.32)	−0.53
White	198/226	208/222	−0.5 (−0.72, −0.33)	−0.51
Non-White	5/6	13/15	−0.1 (−1.17, 0.95)	−0.11
Hispanic/Latino	32/36	30/33	−0.3 (−0.85, 0.17)	−0.33
Not Hispanic/Latino	171/196	191/204	−0.6 (−0.76, −0.34)	−0.53
US	57/75	65/75	−0.5 (−0.85, −0.12)	−0.45
Non-US	146/157	156/162	−0.5 (−0.76, −0.28)	−0.49

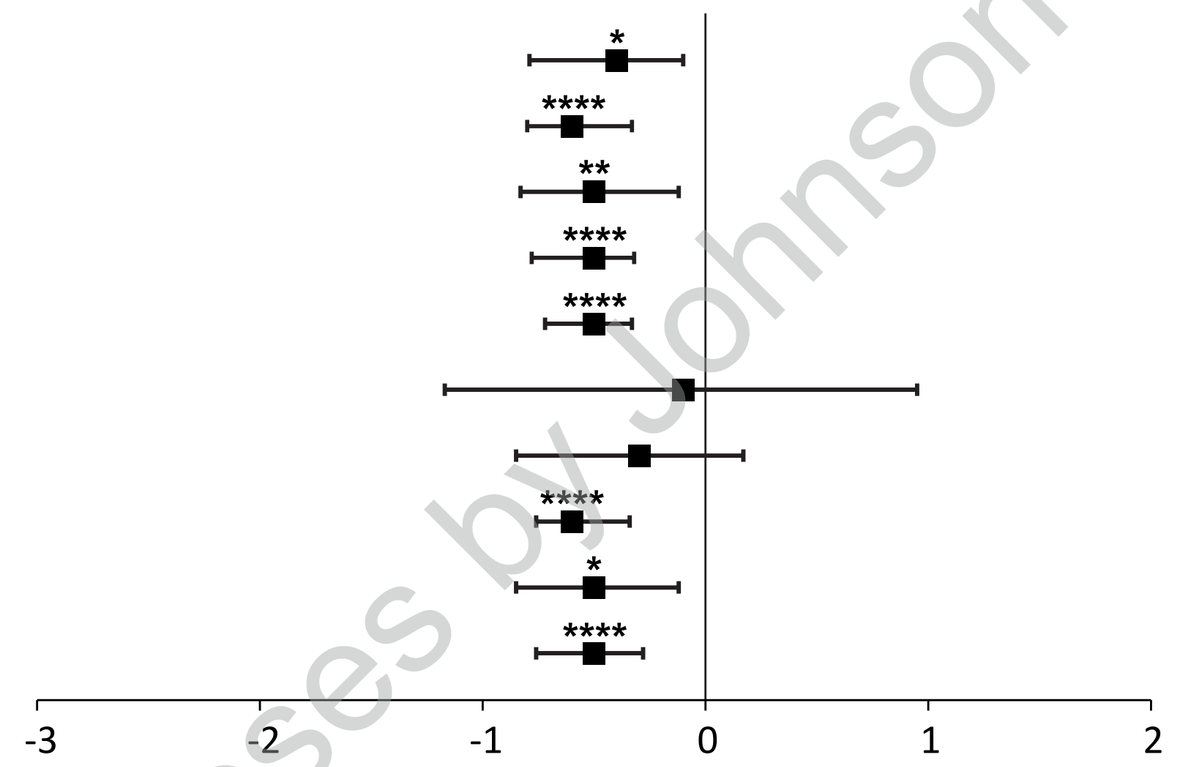
-3 -2 -1 0 1 2
Favors lumateperone + ADT
LSMD (95% CI) vs placebo
Favors placebo + ADT

B. Disease Characteristic Subgroups

	Lumateperone 42 mg + ADT n/N1	Placebo + ADT n/N1	LSMD (95% CI)	Effect Size
Baseline MADRS Total score <32	121/137	118/124	−0.5 (−0.78, −0.27)	−0.51
Baseline MADRS Total score ≥32	82/95	103/113	−0.5 (−0.76, −0.18)	−0.46
SSRI	133/153	131/143	−0.5 (−0.71, −0.22)	−0.45
SNRI or other	70/79	90/94	−0.6 (−0.93, −0.29)	−0.59
1 ADT failure	153/175	166/178	−0.3 (−0.54, −0.10)	−0.32
2 ADT failures	50/57	55/59	−1.1 (−1.53, −0.76)	−1.13
Anxious distress at baseline	79/88	82/91	−0.5 (−0.86, −0.22)	−0.52
No anxious distress at baseline	124/144	139/146	−0.5 (−0.76, −0.27)	−0.50

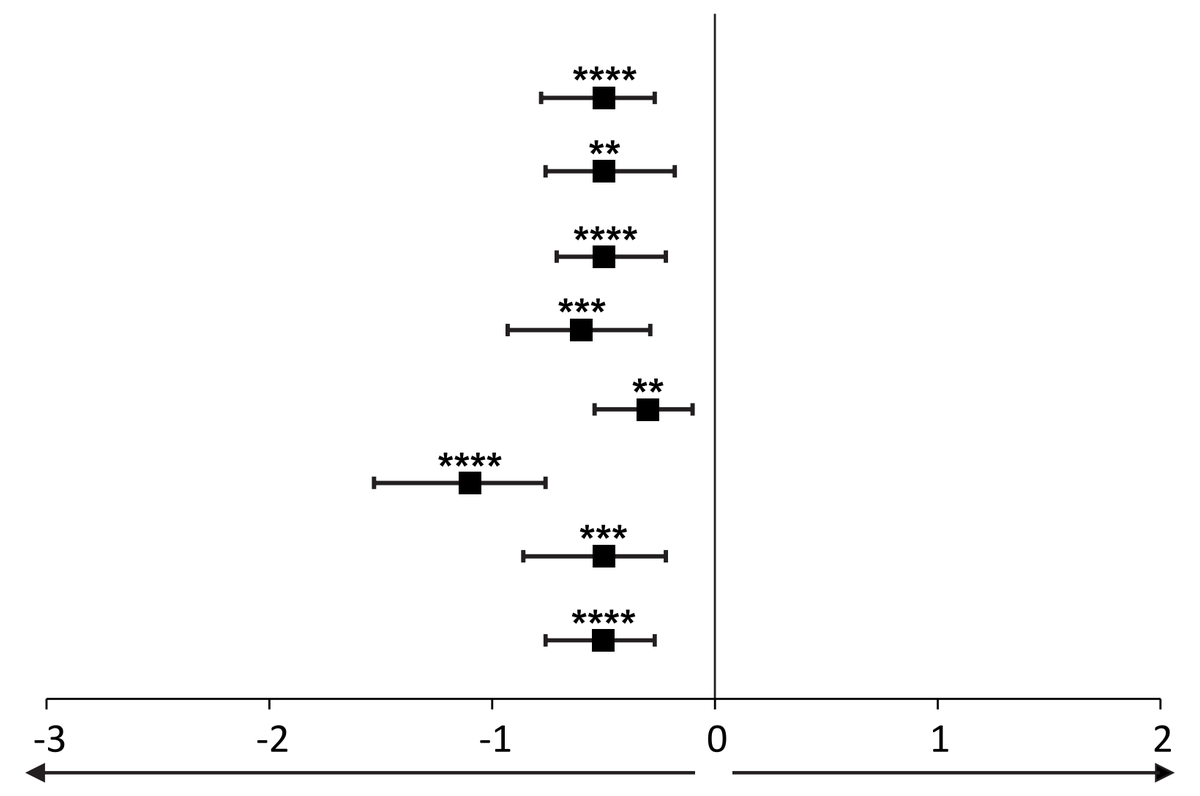
-3 -2 -1 0 1 2
Favors lumateperone + ADT
LSMD (95% CI) vs placebo
Favors placebo + ADT

P*<.05 *P*<.01 ****P*<.001 *****P*<.0001. MMRM in the mITT population.
n=Number of patients with Day 43 assessment; N1=Number of patients in each group at baseline.
ADT, antidepressant therapy; CGI-S, Clinical Global Impression–Severity; LSMD, least squares mean difference; MADRS, Montgomery-Åsberg Depression Rating Scale; mITT, modified intent-to-treat; MMRM, mixed-effects model for repeated measures; SNRI, serotonin-norepinephrine reuptake inhibitor; SSRI, selective serotonin reuptake inhibitor.

Figure 3. Change From Baseline to Day 43 in QIDS-SR-16 Total Score Among (A) Demographic and (B) Disease Characteristic Subgroups (ITT Population)

A. Demographic Subgroups

	Lumateperone 42 mg + ADT n/N1	Placebo + ADT n/N1	LSMD (95% CI)	Effect Size
≤40 years	75/81	71/75	−2.2 (−3.82, −0.52)	−0.44
>40 years	159/161	161/163	−2.3 (−3.35, −1.16)	−0.46
Male	71/73	72/73	−2.1 (−3.73, −0.47)	−0.43
Female	163/169	160/165	−2.3 (−3.36, −1.20)	−0.47
White	227/235	217/223	−2.2 (−3.15, −1.31)	−0.46
Non-White	7/7	15/15	−1.0 (−5.67, 3.57)	−0.21
Hispanic/Latino	36/37	32/33	−1.4 (−3.78, 1.02)	−0.28
Not Hispanic/Latino	198/205	200/205	−2.4 (−3.34, −1.40)	−0.48
US	74/78	72/76	−0.6 (−2.25, 1.00)	−0.12
Non-US	160/164	160/162	−2.9 (−4.02, −1.82)	−0.58

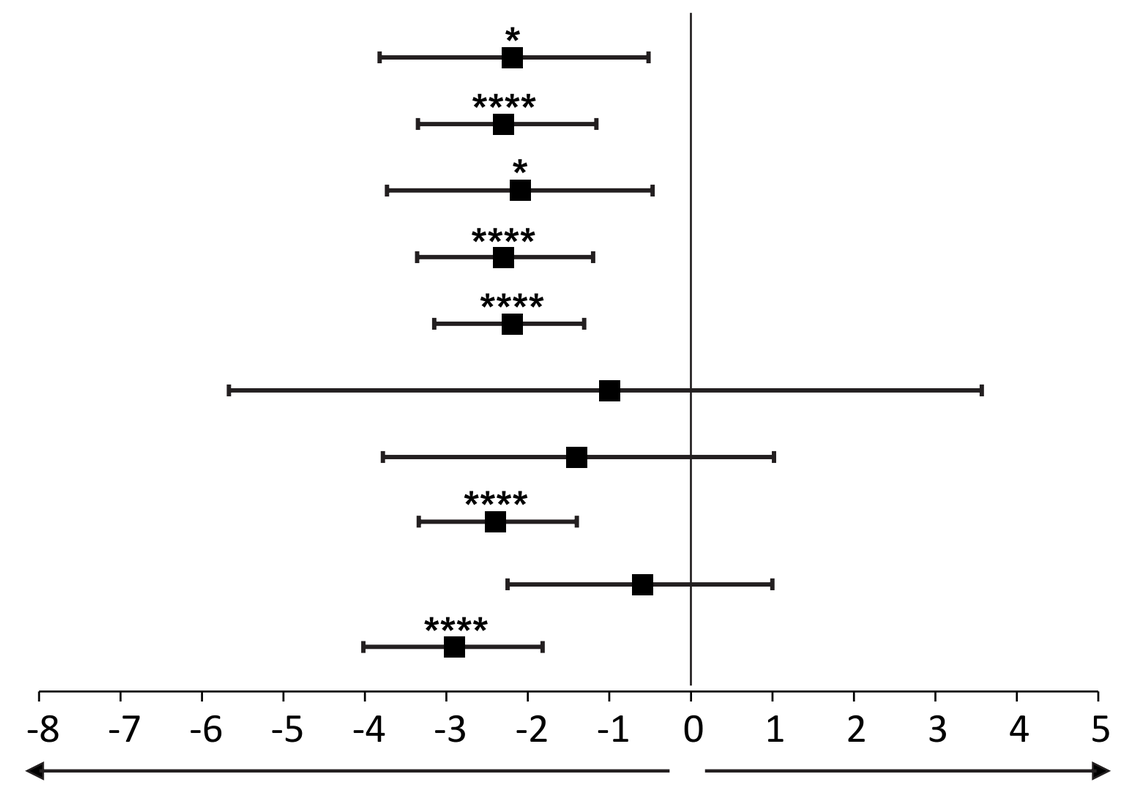
-8 -7 -6 -5 -4 -3 -2 -1 0 1 2 3 4 5
Favors lumateperone + ADT
LSMD (95% CI) vs placebo
Favors placebo + ADT

B. Disease Characteristic Subgroups

	Lumateperone 42 mg + ADT n/N1	Placebo + ADT n/N1	LSMD (95% CI)	Effect Size
Baseline MADRS Total score <32	137/142	122/124	−2.2 (−3.36, −0.96)	−0.45
Baseline MADRS Total score ≥32	97/100	110/114	−2.1 (−3.41, −0.71)	−0.43
SSRI	152/160	138/144	−2.1 (−3.27, −0.97)	−0.43
SNRI or other	82/82	94/94	−2.3 (−3.80, −0.78)	−0.47
1 ADT failure	174/182	173/179	−1.4 (−2.44, −0.37)	−0.29
2 ADT failures	60/60	59/59	−4.6 (−6.37, −2.83)	−0.95
Anxious distress at baseline	90/92	88/91	−2.2 (−3.68, −0.71)	−0.45
No anxious distress at baseline	144/150	144/147	−2.2 (−3.40, −1.09)	−0.46

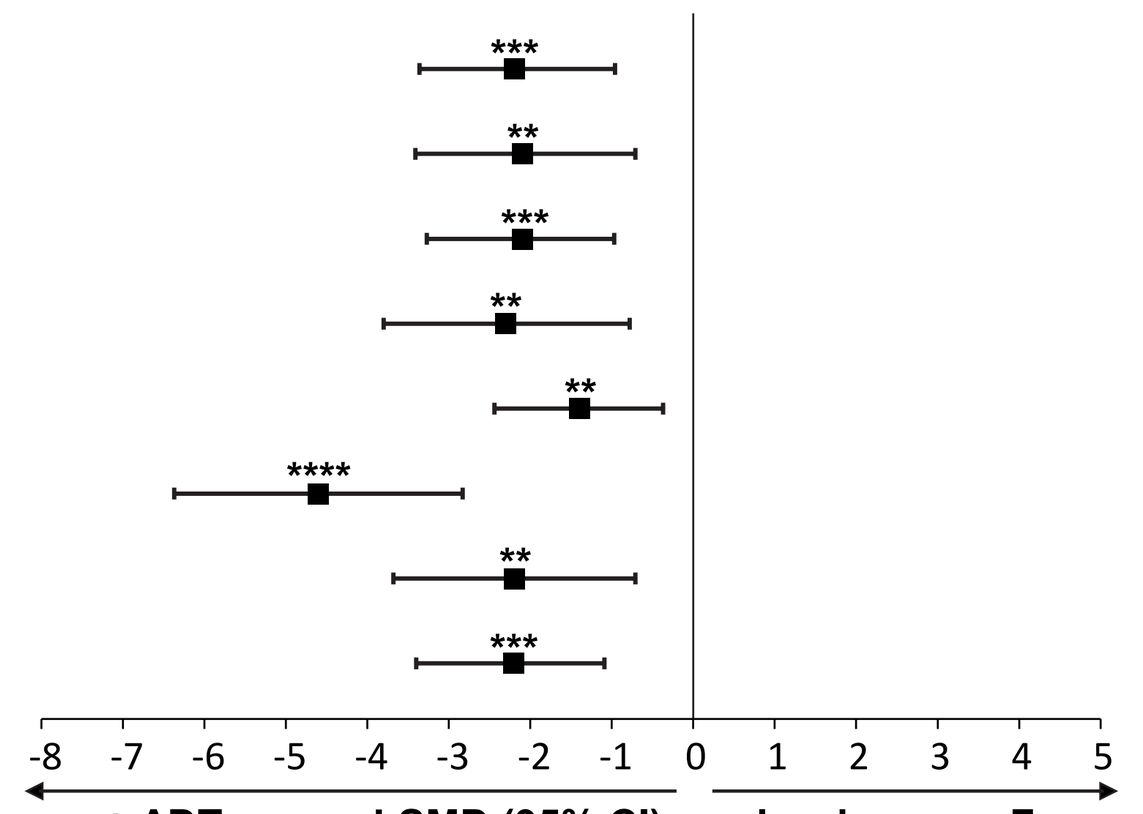
-8 -7 -6 -5 -4 -3 -2 -1 0 1 2 3 4 5
Favors lumateperone + ADT
LSMD (95% CI) vs placebo
Favors placebo + ADT

P*<.05 *P*<.01 ****P*<.001 *****P*<.0001. ANCOVA in the ITT population.
n=Number of patients with Day 43 assessment; N1=Number of patients in each group at baseline.
ADT, antidepressant therapy; ANCOVA, analysis of covariance; LSMD, least squares mean difference; MADRS, Montgomery-Åsberg Depression Rating Scale; ITT, intent-to-treat; QIDS-SR-16, Quick Inventory of Depressive Symptomatology-Self Report-16 item; SNRI, serotonin-norepinephrine reuptake inhibitor; SSRI, selective serotonin reuptake inhibitor.

Conclusions

- Lumateperone 42 mg adjunctive to ADT showed significant improvements over placebo adjunctive to ADT in depression symptoms and disease severity across most subgroups of patients with MDD enrolled in Study 502
- These results further support adjunctive lumateperone 42 mg as an effective option for patients with MDD with inadequate ADT response, regardless of demographic or disease characteristics

Acknowledgments

The authors thank all study investigators, research staff, and patients for their participation. Medical writing support was provided by Joy Steenkamp, PharmD, of Nucleus Global, an Inizio company, funded by Intra-Cellular Therapies, a Johnson & Johnson company.

Disclosures

S Durgam is a former employee of Intra-Cellular Therapies, a Johnson & Johnson company.

R Migliore, WR Earley, Y Mo, and H Lakkis are full-time employees of Intra-Cellular Therapies, a Johnson & Johnson company.

SG Kornstein receives research support from the National Institutes of Health and the National Science Foundation. She has served as a consultant for Lilly, Sage Therapeutics, Reunion Neuroscience, Relmada, AbbVie, Gerbera, and Arrivo BioVentures.

Funding

This study was funded by Intra-Cellular Therapies, a Johnson & Johnson company.

Previously presented at the American Psychiatric Association (APA), May 16-20, 2026, San Francisco, CA, USA.

Neuropsychiatry

Scan the QR code

The QR code is intended to provide scientific information for individual reference, and the information should not be altered or reproduced in any way.

Email questions to Willie R. Earley at wearley@its.jnj.com

Presented at Psych Congress 2026, September 15-19, 2026, New Orleans, LA, USA

References:

1. Yan G, et al. *Psychiatry Res*. 2024;337:115958; 2. Su YA, Si T. *Gen Psychiatry*. 2022;35:e100724; 3. Liu Q, et al. *J Psychiatr Res*. 2020;126:134-140; 4. Rafeyan R, et al. *J Clin Psychiatry*. 2020;81: OT19037BR3; 5. Caplyta (lumateperone). Prescribing information. Intra-Cellular Therapies, Inc; 2026; 6. Titulaer J, et al. *Eur Neuropsychopharmacol*. 2022;62:22-35; 7. Durgam S, et al. *J Clin Psychiatry*. 2025;86:25m15848; 8. Durgam S, et al. *Am J Psychiatry*. 2025;182(12):1072-1082; 9. Durgam S, et al. Poster presented at the 64th Annual Meeting of the American College of Neuropsychopharmacology (ACNP), January 12-15, 2026, Nassau, Bahamas.

This material is distributed for scientific purposes by Johnson & Johnson Innovative Medicine and is not for promotional use.