Efficacy of Adjunctive Lumateperone in Patients With Major Depressive Disorder: A Pooled Post Hoc Analysis of Patients With Clinical Features Associated With a More Challenging Disease Course

Samuel T. Wilkinson, MD[1]; Ibrahim Turkoz, PhD[2]; Brian Curcio, MS[2]; Kristin R. Dorsey, PharmD[2]; Zhiheng Zhang, MS[2]; Christian Edelstein, PharmD[2]

[1]Yale School of Medicine, Department of Psychiatry, New Haven, CT, USA; [2]Johnson & Johnson, Titusville, NJ, USA

118

This material is distributed for scientific purposes by Johnson & Johnson Innovative Medicine and is not for promotional use.

Background

- Major depressive disorder (MDD) is a common and disabling psychiatric condition associated with considerable complexity and substantial burden that can impair functioning and quality of life[1]
 - Some clinical characteristics may be associated with greater disease burden and poorer outcomes in patients with MDD[2]
 - These may include recurrent major depressive episodes (MDEs), longer MDE duration, and history of psychiatric hospitalization[2,3]
 - A significant proportion of patients have suboptimal response to antidepressant therapy (ADT); for these patients, adjunctive treatment may be an option[4-8]
- Lumateperone is FDA approved for the treatment of MDD in adults as an adjunctive therapy to ADT; treatment of schizophrenia in adults; and treatment of depressive episodes associated with bipolar I or II disorder (bipolar depression) in adults, as monotherapy and as an adjunctive therapy with lithium or valproate[9]
 - Lumateperone is a mechanistically novel, multimodal antipsychotic that modulates serotonergic, dopaminergic, and, indirectly, glutamatergic neurotransmission[9,10]
- In 2 Phase 3, randomized, double-blind, placebo-controlled studies (Study 501, NCT04985942; Study 502, NCT05061706), adjunctive lumateperone 42 mg provided superior efficacy versus adjunctive placebo, with meaningful and significant improvements in depressive symptoms (Montgomery-Åsberg Depression Rating Scale [MADRS] Total score), disease severity (Clinical Global Impression–Severity [CGI-S score]), and patient reported outcomes (Quick Inventory of Depressive Symptomatology-Self Report-16 item [QIDS-SR-16])[11,12]
- This pooled analysis of Studies 501 and 502 evaluated the efficacy of adjunctive lumateperone 42 mg across subgroups defined by clinical characteristics associated with more challenging disease course, including a higher number of prior MDEs, longer duration of the current MDE, and prior psychiatric hospitalization

Methods

- Eligible adults (aged 18-65 years) in Studies 501 and 502 met *Diagnostic and Statistical Manual of Mental Disorders, Fifth Edition* (DSM-5) criteria for MDD with inadequate response to 1 or 2 ADT courses (<50% improvement on the Antidepressant Treatment Response Questionnaire) in the current MDE and had MADRS Total score ≥24, CGI-S score ≥4, and QIDS-SR-16 Total score ≥14[11,12]
 - Patients were randomized (1:1) to receive 6 weeks of oral lumateperone 42 mg or placebo per day adjunctive to ADT[11,12]
- Subgroups were defined based on prior number of MDEs (<5 prior MDEs/≥5 prior MDEs), current MDE duration (<6 months/≥6 months), history of psychiatric hospitalization (no/yes), number of ADT treatment failures in the current MDE (1 ADT/2 ADTs), and age of MDD onset (<22 years/≥22 years)
- Mean changes from baseline to end of treatment (EOT) were evaluated in the modified intent-to-treat (mITT) population using mixed-effects model for repeated measures (MMRM; MADRS and CGI-S) or analysis of covariance (ANCOVA; QIDS-SR-16)
- All *P* values were nominal and were not adjusted for multiplicity

Results

Patient Population

- The pooled mITT population comprised 950 patients (lumateperone 42 mg + ADT, n=471; placebo + ADT, n=479)[13]
- Baseline characteristics were generally similar between treatment arms overall (**Table 1**) and within subgroups (**Table 2**)

Table 1. Baseline Demographics and Clinical Characteristics (mITT Population)[13]

	Lumateperone 42 mg + ADT (n=471)	Placebo + ADT (n=479)
Age, mean (range), years	45.2 (18-65)	45.8 (18-65)
Sex, n (%)		
Female	318 (67.5)	325 (67.8)
Male, n (%)	153 (32.5)	154 (32.2)
Race, n (%)		
White	404 (85.8)	413 (86.2)
Asian	41 (8.7)	35 (7.3)
Black	25 (5.3)	24 (5.0)
Other[a]	1 (0.2)	7 (1.5)
Hispanic or Latino ethnicity, n (%)	50 (10.6)	49 (10.2)
Number of lifetime depressive episodes, mean (range)	3.6 (1-30)	4.0 (1-36)
Lifetime history of treatment failures including the current MDE, n (%)		
1	352 (74.7)	352 (73.5)
2	119 (25.3)	127 (26.5)
Prior psychiatric hospitalization, n (%)		
Yes	87 (18.5)	88 (18.4)
No	384 (81.5)	391 (81.6)
MADRS Total score, mean (SD)[b]	30.6 (3.82)	30.8 (3.80)
CGI-S score, mean (SD)[b]	4.7 (0.57)	4.6 (0.57)
QIDS-SR-16 Total score, mean (SD)[c]	18.0 (2.42)	17.8 (2.40)

[a]Other includes Native Hawaiian or other Pacific Islander, multiple, or other. [b]Baseline mean MADRS Total score (lumateperone 42 mg + ADT, 30.6; placebo + ADT, 30.8) and CGI-S score (lumateperone 42 mg + ADT, 4.7; placebo + ADT, 4.6). [c]ITT population (lumateperone 42 mg + ADT n=483, placebo + ADT n=481).
ADT, antidepressant therapy; CGI-S, Clinical Global Impressions–Severity; MADRS, Montgomery–Åsberg Depression Rating Scale; MDE, major depressive episode; mITT, modified intent-to-treat; QIDS-SR-16, Quick Inventory of Depressive Symptomatology-Self Report-16 item.

Table 2. Baseline Demographics and Clinical Characteristics by Subgroup

	Lumateperone 42 mg + ADT	Placebo + ADT	Lumateperone 42 mg + ADT	Placebo + ADT
Number of Prior MDEs	**<5 Prior MDEs**		**≥5 Prior MDEs**	
	n=353	n=352	n=118	n=127
Age, mean (range), years	43.3 (18-65)	44.1 (18-65)	51.1 (23-65)	50.4 (18-65)
Female, n (%)	236 (66.9)	227 (64.5)	82 (69.5)	98 (77.2)
White, n (%)	301 (85.3)	299 (84.9)	103 (87.3)	114 (89.8)
Number of lifetime MDEs, mean (range)	2.5 (1-4)	2.6 (1-4)	6.7 (5-30)	7.8 (5-36)
Duration of current MDE, mean (range), months	7.2 (2-18)	7.0 (2-18)	6.6 (2-18)	6.4 (2-18)
Current MDE Duration	**<6 Months**		**≥6 Months**	
	n=250	n=261	n=221	n=218
Age, mean (range), years	46.1 (18-65)	46.7 (18-65)	44.3 (18-65)	44.8 (18-65)
Female, n (%)	160 (64.0)	177 (67.8)	158 (71.5)	148 (67.9)
White, n (%)	218 (87.2)	232 (88.9)	186 (84.2)	181 (83.0)
Number of lifetime MDEs, mean (range)	3.8 (1-30)	4.0 (1-20)	3.3 (1-11)	4.0 (1-36)
Duration of current MDE, mean (range), months	3.6 (1.8-6.0)	3.6 (1.6-6.0)	10.6 (6.0-17.9)	10.7 (6.1-17.7)
History of Psychiatric Hospitalization	**No**		**Yes**	
	n=384	n=391	n=87	n=88
Age, mean (range), years	44.6 (18-65)	45.1 (18-65)	48.2 (18-65)	49.0 (18-65)
Female, n (%)	263 (68.5)	267 (68.3)	55 (63.2)	58 (65.9)
White, n (%)	323 (84.1)	329 (84.1)	81 (93.1)	84 (95.5)
Number of lifetime MDEs, mean (range)	3.2 (1-15)	3.5 (1-36)	5.2 (1-30)	6.0 (1-20)
Duration of current MDE, mean (range), months	7.4 (1.9-17.8)	7.1 (1.6-17.7)	5.5 (1.8-17.9)	5.6 (1.9-17.5)
Number of ADT Treatment Failures in the Current MDE	**1 ADT Failure**		**2 ADT Failures**	
	n=424	n=418	n=47	n=61
Age, mean (range), years	45.5 (18-65)	46.4 (18-65)	43.1 (18-65)	41.6 (19-64)
Female, n (%)	284 (67.0)	285 (68.2)	34 (72.3)	40 (65.6)
White, n (%)	363 (85.6)	360 (86.1)	41 (87.2)	53 (86.9)
Number of lifetime MDEs, mean (range)	3.6 (1-30)	4.1 (1-36)	3.0 (1-10)	3.2 (1-11)
Duration of current MDE, mean (range), months	6.8 (1.8-17.9)	6.4 (1.6-17.7)	9.4 (3.8-17.2)	10.1 (2.9-17.7)
Age of MDD Onset	**<22 Years**		**≥22 Years**	
	n=81	n=78	n=390	n=401
Age, mean (range), years	33.6 (18-64)	33.7 (18-65)	47.7 (23-65)	48.2 (22-65)
Female, n (%)	56 (69.1)	56 (71.8)	262 (67.2)	269 (67.1)
White, n (%)	69 (85.2)	62 (79.5)	335 (85.9)	351 (87.5)
Number of lifetime MDEs, mean (range)	3.6 (1-11)	4.3 (1-20)	3.6 (1-30)	3.9 (1-36)
Duration of current MDE, mean (range), months	8.6 (2.0-17.2)	8.8 (2.6-17.4)	6.7 (1.8-17.9)	6.5 (1.6-17.7)

ADT, antidepressant therapy; MDD, major depressive disorder; MDE, major depressive episode.

Number of Prior MDEs

- Lumateperone 42 mg + ADT versus placebo + ADT significantly improved MADRS Total score, CGI-S score, and QIDS-SR-16 Total score from baseline to EOT regardless of the number of prior MDEs:
 - MADRS Total score least squares mean difference versus placebo (LSMD): <5 prior MDEs, −4.2; ≥5 prior MDEs, −5.7; both *P*<.0001 (**Figure 1**)
 - CGI-S score LSMD: <5 prior MDEs, −0.6; ≥5 prior MDEs, −0.7; both *P*<.0001 (**Figure 2**)
 - QIDS-SR-16 LSMD: <5 prior MDEs, −2.1; ≥5 prior MDEs, −3.1; both *P*<.001 (**Figure 3**)

Current MDE Duration

- Lumateperone 42 mg + ADT versus placebo + ADT significantly improved MADRS Total score, CGI-S score, and QIDS-SR-16 Total score from baseline to EOT both for patients with current MDE duration of <6 months and for those with current MDE duration ≥6 months
 - MADRS Total score LSMD: <6 months, −4.3; ≥6 months, −5.0; both *P*<.0001 (**Figure 1**)
 - CGI-S score LSMD: <6 months, −0.6; ≥6 months, −0.7; both *P*<.0001 (**Figure 2**)
 - QIDS-SR-16 LSMD: <6 months, −2.5; ≥6 months, −2.2; both *P*<.001 (**Figure 3**)

History of Psychiatric Hospitalization

- Similar results were observed for subgroups with and without prior psychiatric hospitalization
 - MADRS Total score LSMD: no, −4.2; yes, −6.3; both *P*<.0001 (**Figure 1**)
 - CGI-S score LSMD: no, −0.6; yes, −0.7; both *P*<.0001 (**Figure 2**)
 - QIDS-SR-16 Total score LSMD: no, −2.1; yes, −3.6; both *P*<.001 (**Figure 3**)

Number of ADT Treatment Failures in the Current MDE

- Lumateperone 42 mg + ADT versus placebo + ADT significantly improved depression severity from baseline to EOT for patients with 1 or 2 ADT treatment failures in the current MDE
 - MADRS Total score LSMD: 1 ADT failure, −4.5; 2 ADT failures, −5.7; both *P*<.001 (**Figure 1**)
 - CGI-S score LSMD: 1 ADT failure, −0.6; 2 ADT failures, −0.9; both *P*<.001 (**Figure 2**)
 - QIDS-SR-16 Total score LSMD: 1 ADT failure, −2.1; 2 ADT failures, −4.4; both *P*<.001 (**Figure 3**)

Age of MDD Onset

- Lumateperone 42 mg + ADT versus placebo + ADT improved MADRS Total score, CGI-S score, and QIDS-SR-16 Total score from baseline to EOT for patients with age of MDD onset <22 years and for those with age of MDD onset ≥22 years; all comparisons were significant except for QIDS-SR-16 Total score for those with MDD onset <22 years of age
 - MADRS Total score LSMD: onset <22 years of age, −4.5 (*P*<.01); MDD onset ≥22 years of age, −4.7 (*P*<.001) (**Figure 1**)
 - CGI-S score LSMD: onset <22 years of age, −0.6; MDD onset ≥22 years of age, −0.6; both *P*<.001 (**Figure 2**)
 - QIDS-SR-16 Total score LSMD: MDD onset <22 years of age, −1.4 (*P*=.064); MDD onset ≥22 years of age, −2.6 (*P*<.001) (**Figure 3**)

Figure 1. Changes From Baseline to EOT in MADRS Total Score by Subgroup

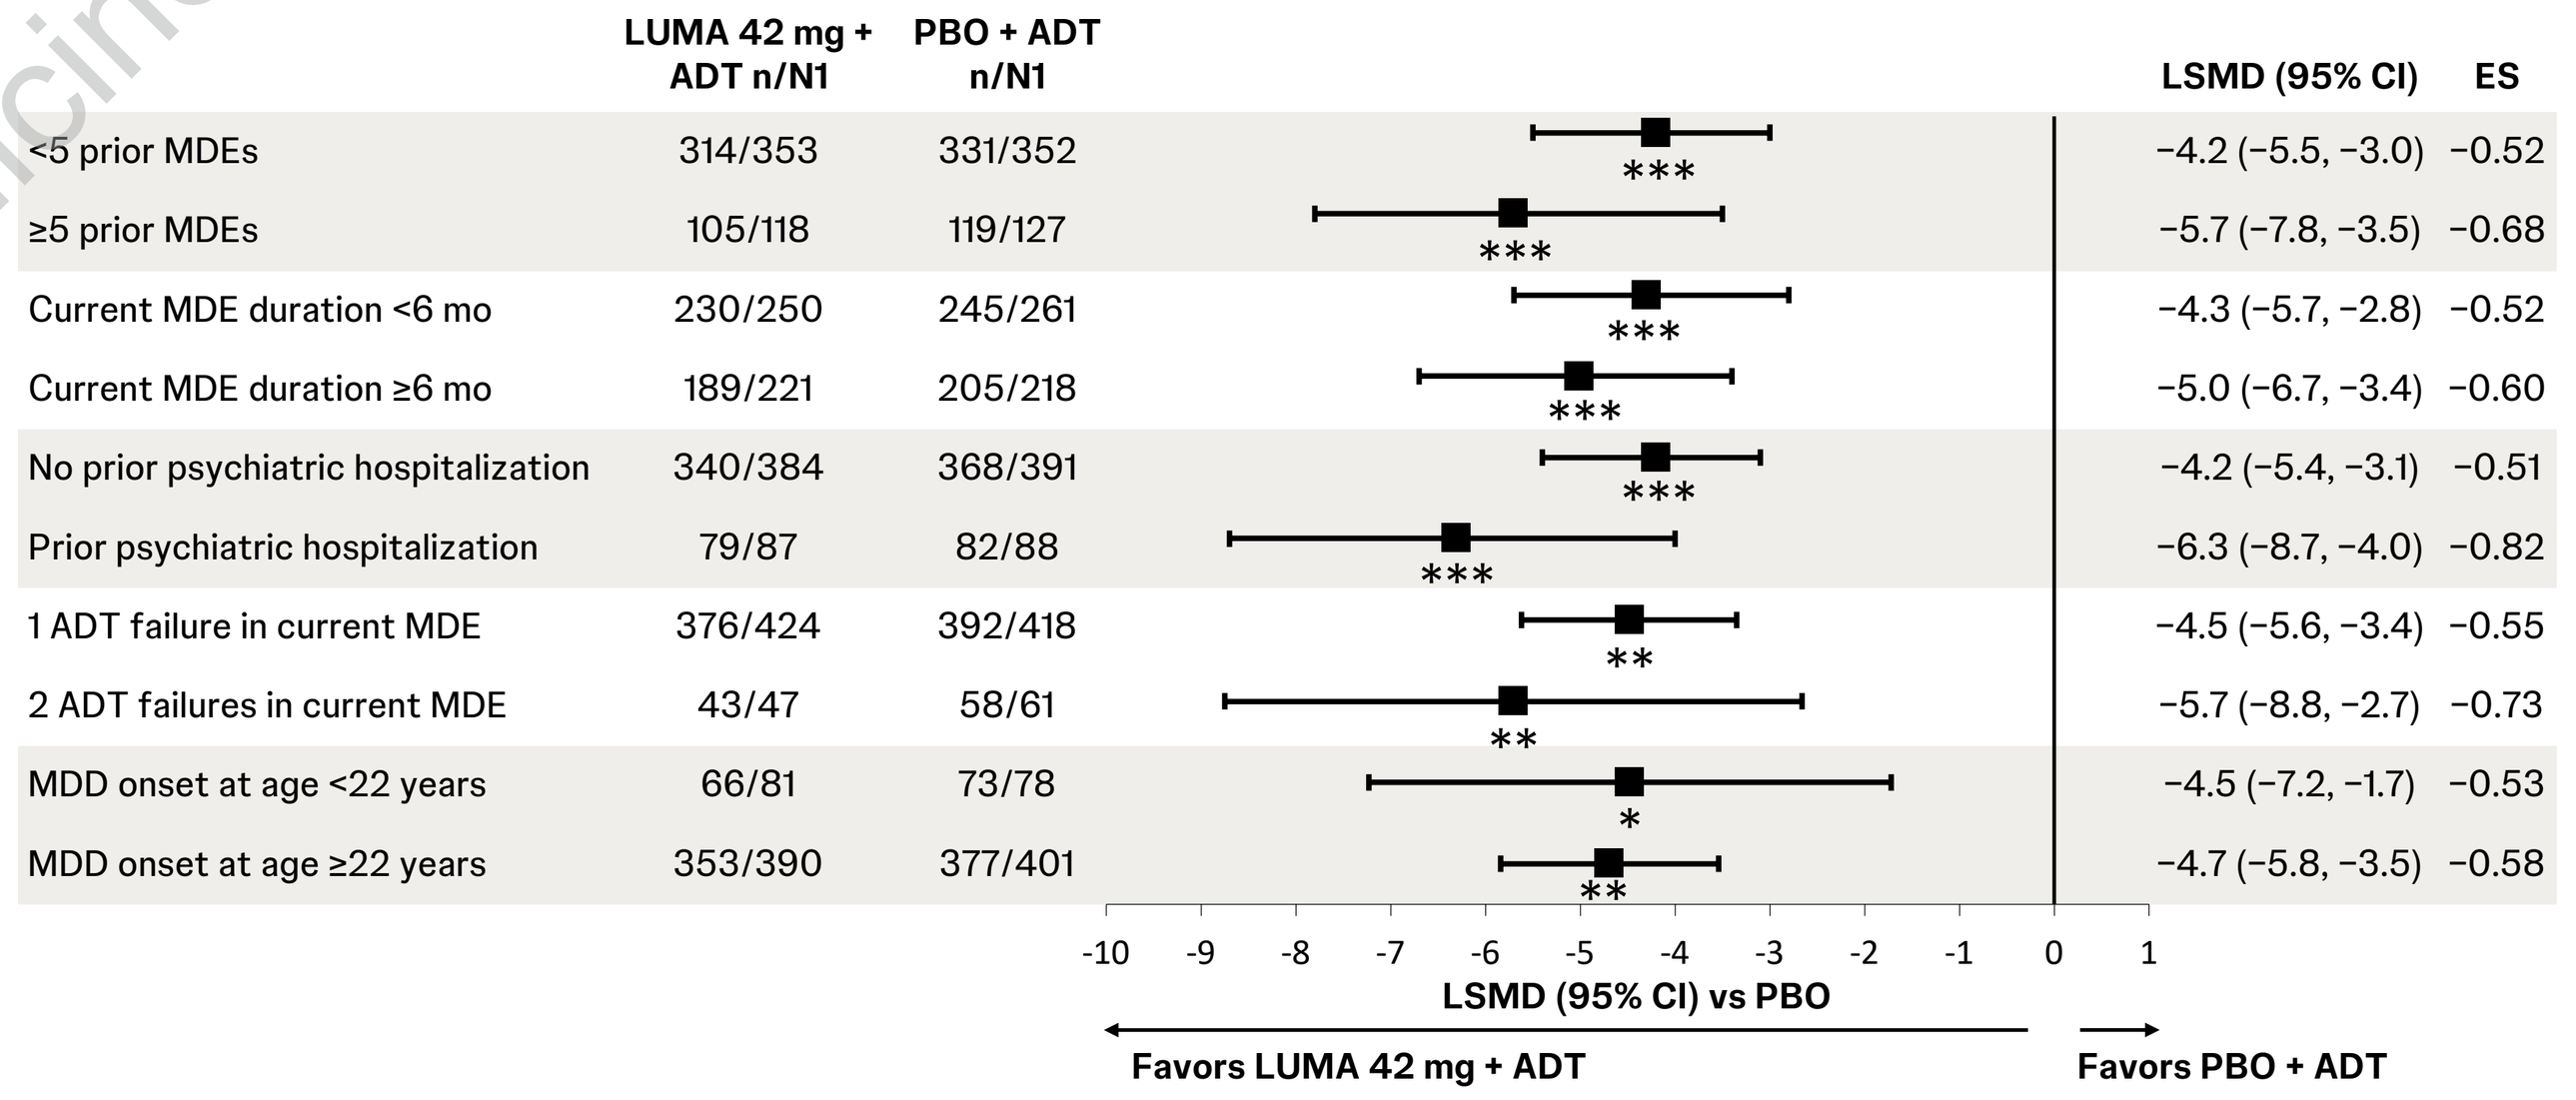
LUMA 42 mg + ADT n/N1
PBO + ADT n/N1
LSMD (95% CI)
ES
<5 prior MDEs 314/353 331/352 −4.2 (−5.5, −3.0) −0.52
≥5 prior MDEs 105/118 119/127 −5.7 (−7.8, −3.5) −0.68
Current MDE duration <6 mo 230/250 245/261 −4.3 (−5.7, −2.8) −0.52
Current MDE duration ≥6 mo 189/221 205/218 −5.0 (−6.7, −3.4) −0.60
No prior psychiatric hospitalization 340/384 368/391 −4.2 (−5.4, −3.1) −0.51
Prior psychiatric hospitalization 79/87 82/88 −6.3 (−8.7, −4.0) −0.82
1 ADT failure in current MDE 376/424 392/418 −4.5 (−5.6, −3.4) −0.55
2 ADT failures in current MDE 43/47 58/61 −5.7 (−8.8, −2.7) −0.73
MDD onset at age <22 years 66/81 73/78 −4.5 (−7.2, −1.7) −0.53
MDD onset at age ≥22 years 353/390 377/401 −4.7 (−5.8, −3.5) −0.58
-10 -9 -8 -7 -6 -5 -4 -3 -2 -1 0 1
LSMD (95% CI) vs PBO
Favors LUMA 42 mg + ADT
Favors PBO + ADT

* *P*<.01 ** *P*<.001 *** *P*<.0001. Mixed-effects model for repeated measures in the mITT population. ADT, antidepressant therapy; EOT, end of treatment; ES, effect size; LSMD, least squares mean difference; LUMA, lumateperone; MADRS, Montgomery–Asberg Depression Rating Scale; MDD, major depressive disorder; MDE, major depressive episode; mITT, modified intent-to-treat; n, number of patients with Day 43 assessment; N1, number of mITT patients in each group at baseline; PBO, placebo.

Figure 2. Changes From Baseline to EOT in CGI-S Score by Subgroup

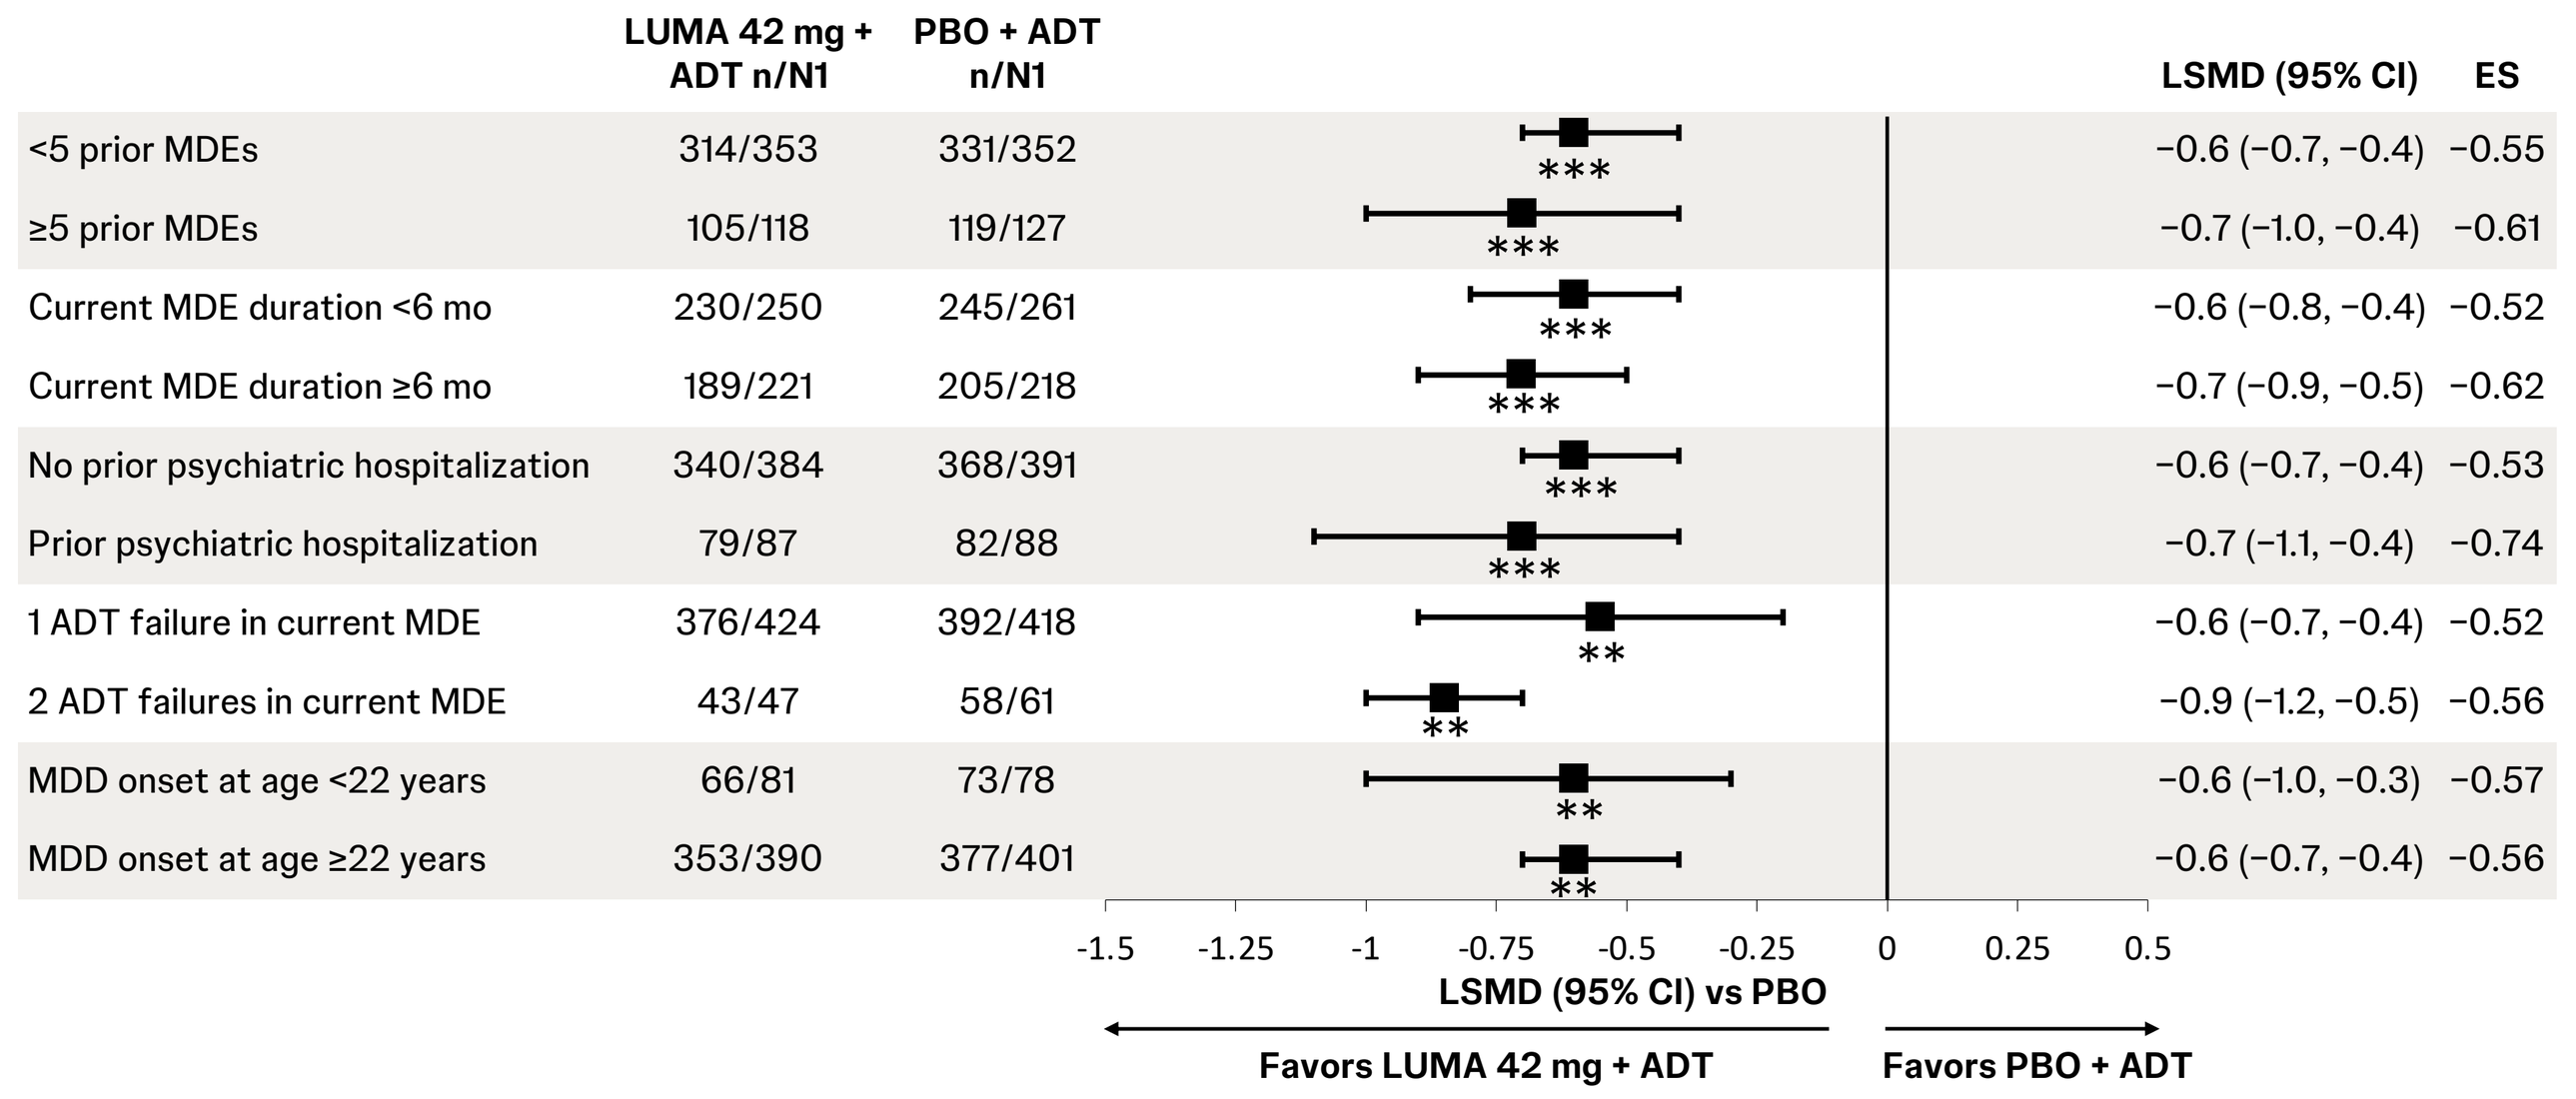
LUMA 42 mg + ADT n/N1
PBO + ADT n/N1
LSMD (95% CI)
ES
<5 prior MDEs 314/353 331/352 −0.6 (−0.7, −0.4) −0.55
≥5 prior MDEs 105/118 119/127 −0.7 (−1.0, −0.4) −0.61
Current MDE duration <6 mo 230/250 245/261 −0.6 (−0.8, −0.4) −0.52
Current MDE duration ≥6 mo 189/221 205/218 −0.7 (−0.9, −0.5) −0.62
No prior psychiatric hospitalization 340/384 368/391 −0.6 (−0.7, −0.4) −0.53
Prior psychiatric hospitalization 79/87 82/88 −0.7 (−1.1, −0.4) −0.74
1 ADT failure in current MDE 376/424 392/418 −0.6 (−0.7, −0.4) −0.52
2 ADT failures in current MDE 43/47 58/61 −0.9 (−1.2, −0.5) −0.56
MDD onset at age <22 years 66/81 73/78 −0.6 (−1.0, −0.3) −0.57
MDD onset at age ≥22 years 353/390 377/401 −0.6 (−0.7, −0.4) −0.56
-1.5 -1.25 -1 -0.75 -0.5 -0.25 0 0.25 0.5
LSMD (95% CI) vs PBO
Favors LUMA 42 mg + ADT
Favors PBO + ADT

** *P*<.001 *** *P*<.0001. Mixed-effects model for repeated measures in the mITT population. ADT, antidepressant therapy; CGI-S, Clinical Global Impressions–Severity; EOT, end of treatment; ES, effect size; LSMD, least squares mean difference; LUMA, lumateperone; MDD, major depressive disorder; MDE, major depressive episode; mITT, modified intent-to-treat; n, number of patients with Day 43 assessment; N1, number of mITT patients in each group at baseline; PBO, placebo.

Figure 3. Changes From Baseline to EOT in QIDS-SR-16 Total Score by Subgroup

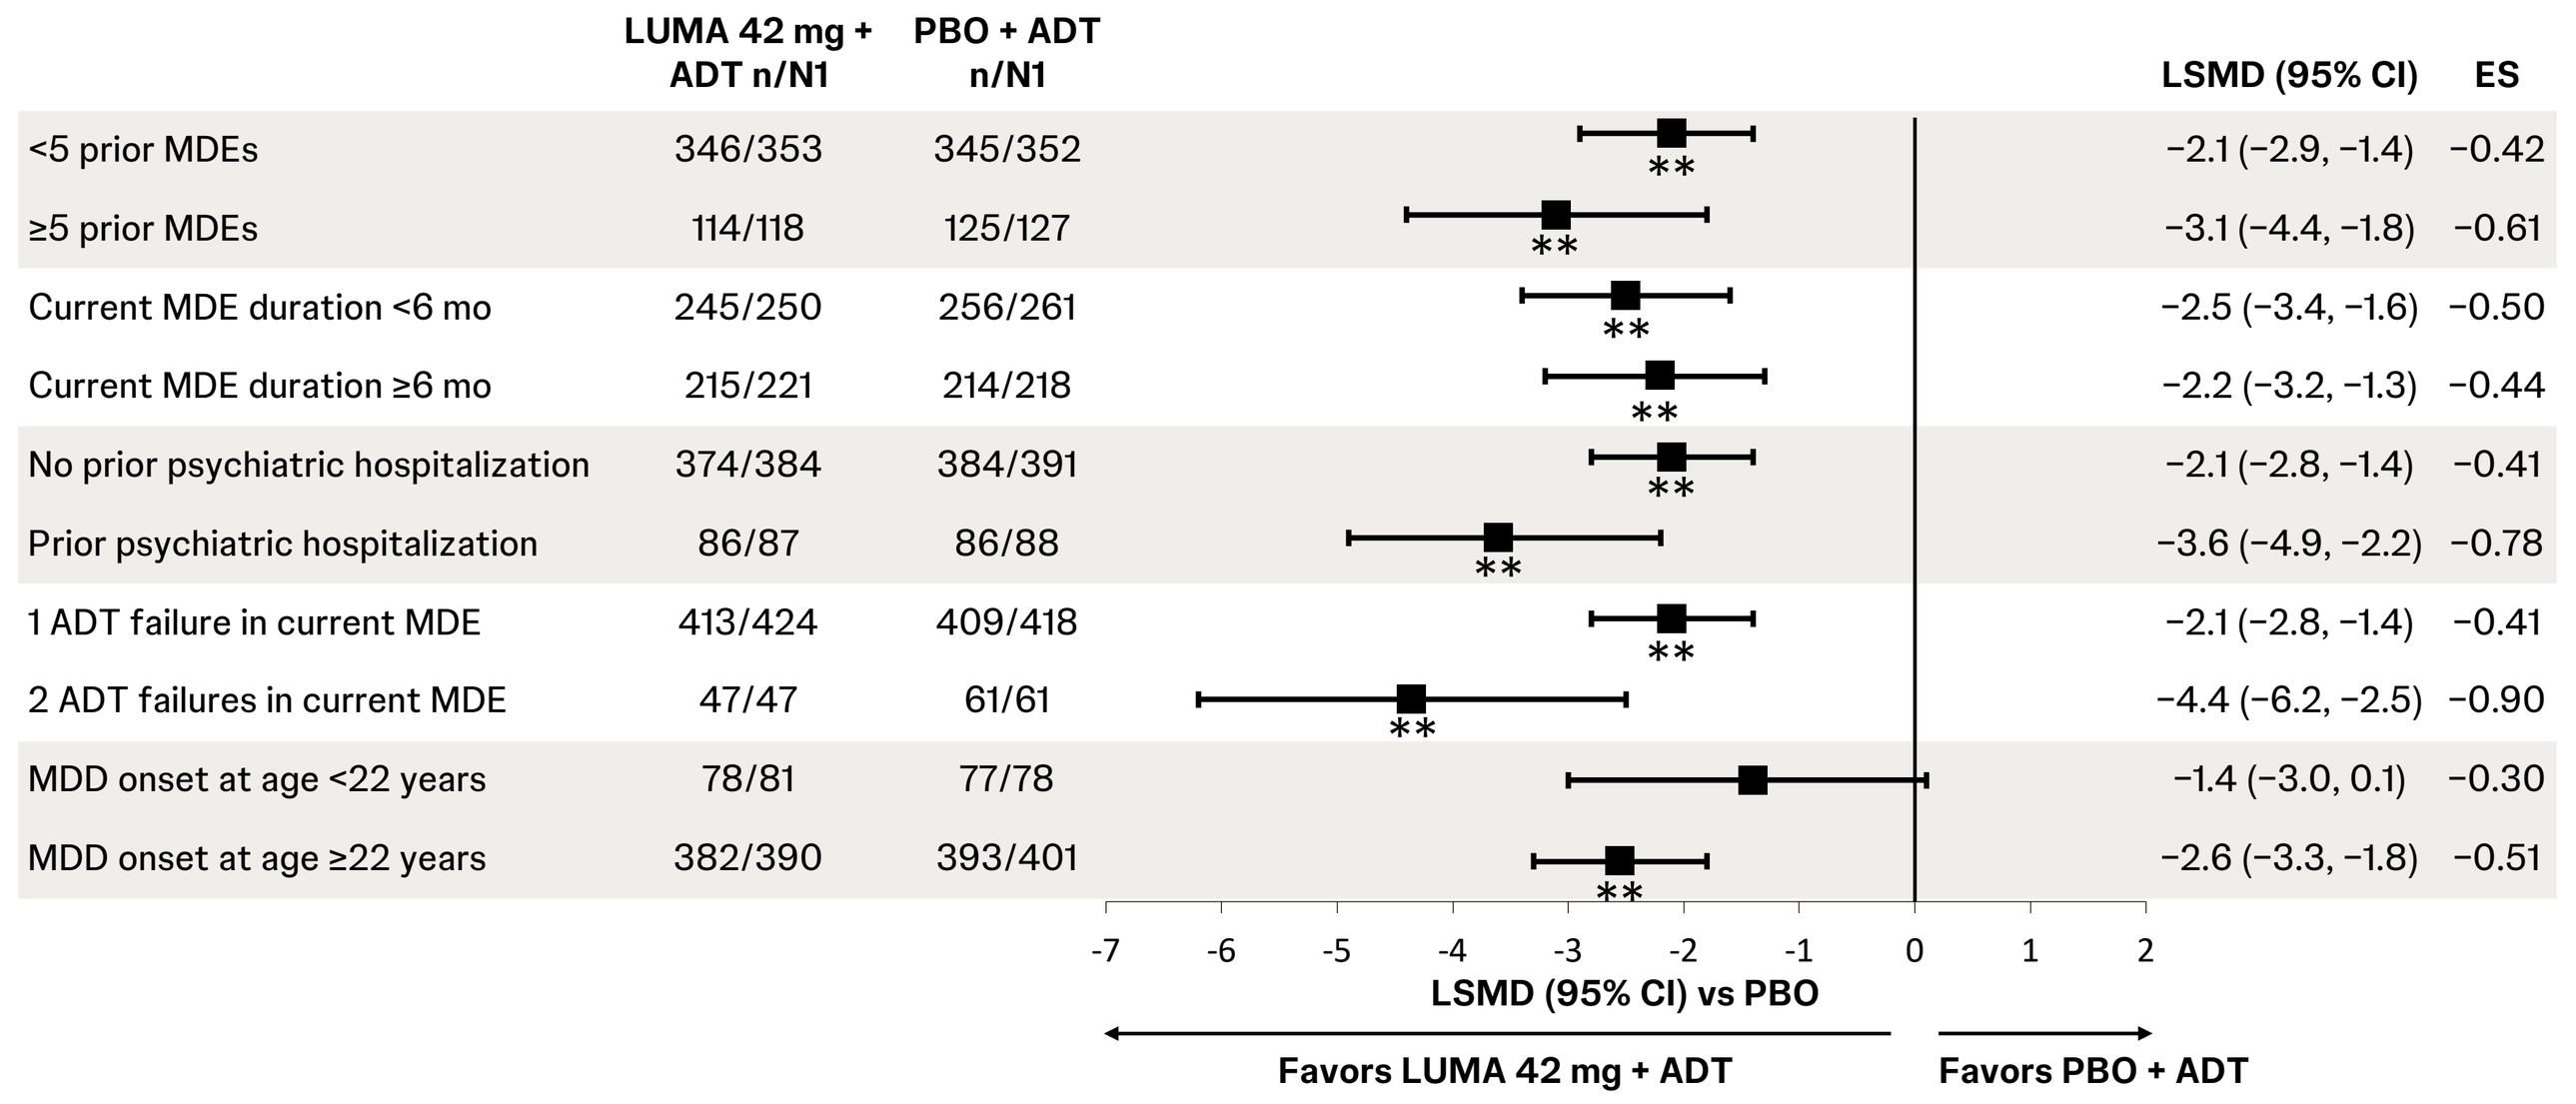
LUMA 42 mg + ADT n/N1
PBO + ADT n/N1
LSMD (95% CI)
ES
<5 prior MDEs 346/353 345/352 −2.1 (−2.9, −1.4) −0.42
≥5 prior MDEs 114/118 125/127 −3.1 (−4.4, −1.8) −0.61
Current MDE duration <6 mo 245/250 256/261 −2.5 (−3.4, −1.6) −0.50
Current MDE duration ≥6 mo 215/221 214/218 −2.2 (−3.2, −1.3) −0.44
No prior psychiatric hospitalization 374/384 384/391 −2.1 (−2.8, −1.4) −0.41
Prior psychiatric hospitalization 86/87 86/88 −3.6 (−4.9, −2.2) −0.78
1 ADT failure in current MDE 413/424 409/418 −2.1 (−2.8, −1.4) −0.41
2 ADT failures in current MDE 47/47 61/61 −4.4 (−6.2, −2.5) −0.90
MDD onset at age <22 years 78/81 77/78 −1.4 (−3.0, 0.1) −0.30
MDD onset at age ≥22 years 382/390 393/401 −2.6 (−3.3, −1.8) −0.51
-7 -6 -5 -4 -3 -2 -1 0 1 2
LSMD (95% CI) vs PBO
Favors LUMA 42 mg + ADT
Favors PBO + ADT

** *P*<.001. Analysis of covariance in the mITT population. ADT, antidepressant therapy; EOT, end of treatment; ES, effect size; LSMD, least squares mean difference; LUMA, lumateperone; MDD, major depressive disorder; MDE, major depressive episode; mITT, modified intent-to-treat; n, number of patients with Day 43 assessment; N1, number of mITT patients in each group at baseline; PBO, placebo; QIDS-SR-16, Quick Inventory of Depressive Symptomatology-Self Report-16 item.

Conclusions

- Lumateperone 42 mg + ADT significantly improved MADRS Total score, CGI-S score, and QIDS-SR-16 Total score versus placebo + ADT across subgroups defined by prior number of MDEs (<5 prior MDEs and ≥5 prior MDEs), current MDE duration (<6 months and ≥6 months), history of psychiatric hospitalization (no and yes), number of ADT treatment failures in the current MDE (1 ADT and 2 ADTs), and age of MDD onset (≥22 years)
 - Significant improvements in MADRS Total score and CGI-S score were also observed in the subgroup with age of MDD onset <22 years
- These findings support adjunctive lumateperone 42 mg as an effective treatment option for patients with MDD with high or low clinical burden, both early- and late-illness patients, including those with clinical features associated with a more challenging disease course

Acknowledgments

The authors thank all study investigators, research staff, and patients for their participation. Medical writing support was provided by JoAnna Anderson, PhD, CMPP, of Nucleus Global, an Inizio company, funded by Intra-Cellular Therapies, a Johnson & Johnson company.

Disclosures

This study was sponsored by Johnson & Johnson.

ST Wilkinson has received contract research funding (administered through Yale University) from Neurocrine Biosciences, NRX Pharmaceuticals, and Oui Therapeutics. He has served as a consultant or provided data safety and monitoring board services for Atai-Beckley, Definium Therapeutics, EPIC, Joyous, LivaNova, Neumora, Osmind, Sage Therapeutics, and Sooma Medical.

I Turkoz, B Curcio, KR Dorsey, Z Zhang, and C Edelstein are employees of Johnson & Johnson.

Neuropsychiatry

Scan the QR code
The QR code is intended to provide scientific information for individual reference, and the information should not be altered or reproduced in any way

Email questions to Kristin R. Dorsey at kdorsey@its.jnj.com

Presented at Psych Congress 2026, September 15-19, 2026, New Orleans, LA, USA

References:

1. Earley WR, et al. *CNS Drugs*. 2026; doi: 10.1007/s40263-026-01314-8 [Published online July 15, 2026]; 2. Costa T, et al. *J Psychopharmacol*. 2022;36:545-56; 3. McAllister-Williams RH, et al. *J Affect Disord*. 2020;267:264-82; 4. Rush AJ, et al. *Am J Psychiatry*. 2006;163:1905-17; 5. Trivedi MH, et al. *Am J Psychiatry*. 2006;163:28-40; 6. Gelenberg A, et al. 2010. Accessed on: August 11, 2026. https://psychiatryonline.org/pb/assets/raw/sitewide/practice_guidelines/guidelines/mdd.pdf; 7. National Institute for Health and Care Excellence. 2022. Accessed on: August 11, 2026. https://www.nice.org.uk/guidance/ng222; 8. Rafeyan R, et al. *J Clin Psychiatry*. 2020;81:27570; 9. Caplyta (lumateperone). Prescribing information. Intra-Cellular Therapies, Inc; 2026; 10. Davis RE, et al. *Expert Rev Neurother*. 2016;16:601-14; 11. Durgam S, et al. *J Clin Psychiatry*. 2025;86:25m15848; 12. Durgam S, et al. *Am J Psychiatry*. 2025;182:1072-82; 13. Durgam S, et al. Presented at the 36th World Congress of Neuropsychopharmacology/9th AsCNP Congress (CINP-AsCNP) Joint Congress, June 15-18, 2025, Melbourne, Australia.