40

Assessment of Lumateperone Partial Agonist Activity at Presynaptic Dopamine D_2 Autoreceptors: Reversal of Haloperidol-Induced Tyrosine Hydroxylase Phosphorylation in Mouse Striatum

Maxine Chen, PhD[1]; Gretchen L. Snyder, PhD[1]; Sophie Dutheil, PhD[1]

[1]Johnson & Johnson, Titusville, NJ, USA

Background

- Lumateperone is FDA approved for the treatment of schizophrenia in adults, for the treatment of depressive episodes associated with bipolar I or II disorder (bipolar depression) in adults as monotherapy and as adjunctive therapy with lithium or valproate, and as an adjunctive treatment for major depressive disorder[1]
- Lumateperone is a mechanistically novel, multimodal antipsychotic that modulates serotonergic and dopaminergic neurotransmission, and indirectly modulates glutamatergic neurotransmission[2]
 - Lumateperone has high binding affinity for serotonin 5-HT_{2A} receptors (K_i=0.54 nM) and moderate binding affinity for dopamine D_2 receptors (K_i=32 nM)[1]; D_2 receptor occupancy of lumateperone (39%) is lower than that of other antipsychotics (65%-80%)[3]
- Most available antipsychotics act in part as D_2 antagonists (eg, haloperidol, quetiapine, lurasidone)[4,5] or as partial D_2 agonists (eg, aripiprazole, brexpiprazole, cariprazine)[6] (**Figure 1**)
 - While D_2 receptor blockade is essential for antipsychotic activity, extrapyramidal symptoms (EPS) are observed with high occupancy from D_2 receptor antagonists[3,4]
- The novel mechanism of lumateperone, including presynaptic partial agonism while maintaining postsynaptic D_2 blockade with low D_2 receptor occupancy, balances out the dopaminergic tone and may contribute to the low risk of EPS and akathisia with lumateperone[3,7]
- Tyrosine hydroxylase (TH) is the rate-limiting enzyme in dopamine synthesis, and phosphorylation at serine 40 (S40) is essential for its catalytic activity to drive dopamine production[8,9]
 - In vivo, treatment with the D_2 receptor antagonist haloperidol increases the levels of phosphorylated TH (p-TH) at residue S40 in the mouse striatum[4,8]
- This analysis provides evidence for the presynaptic partial agonism of lumateperone at D_2 receptors, which is a key contributor to the favorable safety profile observed with lumateperone in clinical trials[10]
- The hypothesis investigated here was that lumateperone acting as a presynaptic partial agonist would reduce the levels of haloperidol-induced p-TH in mouse striatum

Figure 1 . Effects of Dopamine D_2 Receptor Antagonist (A) and Agonist (B) on Dopamine Biosynthesis Through Modulation of TH Phosphorylation

Panel A: Antagonist binding (1) blocks the normal inhibition of AC activity (2), resulting in an increase in formation of cAMPc, activation of PKA (3) and phosphorylation of TH at S40 (4), which promotes conversion of tyrosine to L-DOPA and dopamine biosynthesis rate (5).

Panel B: Agonist binding (1) enhances the normal inhibition of AC activity by the D_2 receptor (2), resulting in a decrease in formation of cAMPc, reduced PKA activity (3) and reduced phosphorylation of TH at S40 (4), leading to a reduction in the driving force for conversion of tyrosine to L-DOPA and dopamine biosynthesis rate (5).

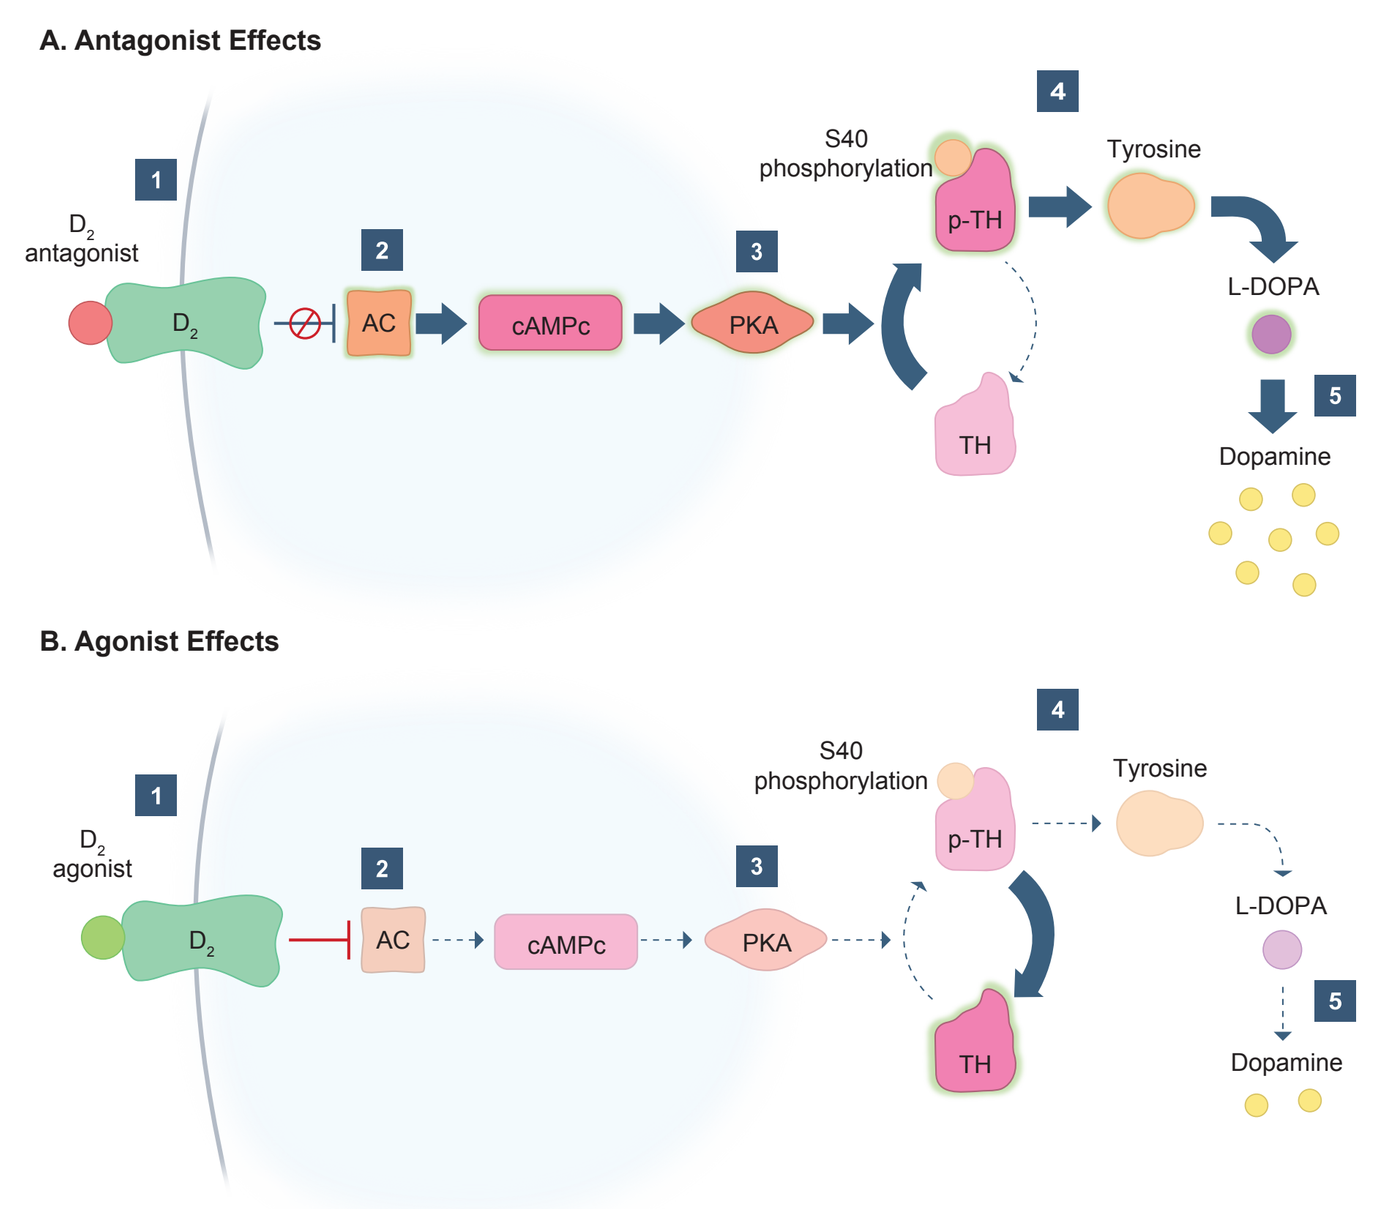
A. Antagonist Effects
D_2 antagonist
D_2
AC
cAMPc
PKA
S40 phosphorylation
p-TH
TH
Tyrosine
L-DOPA
Dopamine
B. Agonist Effects
D_2 agonist
D_2
AC
cAMPc
PKA
S40 phosphorylation
p-TH
TH
Tyrosine
L-DOPA
Dopamine

AC, adenylyl cyclase; cAMPc, cyclic adenosine monophosphate (cat); p-TH, phosphorylated TH; PKA, protein kinase A; S40, serine 40; TH, tyrosine hydroxylase.

Methods

Animals and Treatments

- Adult C57BL/6 male mice were co-administered an intraperitoneal injection of the D_2 antagonist drug haloperidol (1 mg/kg)[7] or its vehicle with one of the following:

Full dopamine D_2 receptor agonist ⇨	Quinpirole hydrochloride (1 or 2 mg/kg)
D_2 partial agonists ⇨	Aripiprazole (3 mg/kg)[7] Cariprazine (3 mg/kg)[11] Brexpiprazole (10 mg/kg)[12] Lumateperone (6 mg/kg)[7]
D_2 receptor antagonists ⇨	Lurasidone hydrochloride (1 or 3 mg/kg)[5] Quetiapine fumerate (5 or 10 mg/kg)[7]

Striatal Tissue Collection

- One hour after drug treatment, mice were humanely euthanized using focused microwave irradiation of the head (Muromachi Kikai)
- Striatal tissue was bilaterally dissected and snap frozen in liquid nitrogen

Western Blotting

- BCA assay (Pierce, Thermo Fisher) was used to quantify the protein concentration of each
- Normalized striatal samples (20 μg/μL) were separated on 2%-14% Bis-Tris gradient protein gels, running at 140 volts for 75 min
- Proteins were then transferred to nitrocellulose membranes for analysis

Data Analysis

- Data were analyzed by 1-way analysis of variance (ANOVA) using GraphPad Prism 9 software, and results are presented as mean ± SEM
- Each treatment group was normalized to the control group
- Analysis was followed by a post hoc pairwise multiple comparison procedure using the Bonferroni method if the interaction was significant (P<.05)
- Outliers were removed using the median absolute deviation (MAD) equation (median ± 2.5× the MAD method for outlier detection), which is a more robust measure of dispersion than the mean ± 2 or 3 standard deviation[13]

Table 1. Drugs and Antibodies

Drugs	Manufacturer
• Lumateperone (ITI-007)	As a tosylate salt, provided by the Medicinal Chemistry group at Intra-Cellular Therapies, a Johnson & Johnson company (lot #13337075)
• Haloperidol (H1512) • Lurasidone hydrochloride (#SML2069)	Obtained from Sigma-Aldrich
• (-) Quinpirole hydrochloride (#1061) • Aripiprazole (#5584) • Quetiapine hemifumerate (#4735)	Obtained from BioTechne (Tocris)
• Brexpiprazole	Obtained from Advanced Chemblocks (#I-9700)
• Cariprazine hydrochloride	Obtained from MedChemExpress (#HY-14763A)
Antibodies	**Manufacturer**
• Phospho-tyrosine hydroxylase (Ser40)	Obtained from Cell Signaling (#2791S); dilution 1:750
• Total anti-tyrosine hydroxylase antibody	Obtained from Abcam (ab76442); dilution 1:2000
• Loading control: GAPDH, vinculin, or β-actin	Obtained from Cell Signaling: GAPDH (#2118; 1:15,000), vinculin (#13901; 1:2000), β-actin (#4967; (1:15,000)

Results

- In mouse striatum, the D_2 antagonist haloperidol (which blocks D_2 receptors on striatal dopamine terminals) significantly increased dopamine response, inducing an ≈3-fold increase in p-TH at the regulatory S40 residue (P<.001) (**Figure 2A**)
 - Co-injection of haloperidol with lumateperone 3 mg/kg (P<.01) or 6 mg/kg (P<.001) significantly reduced p-TH levels compared with haloperidol alone
 - A representative Western blot of striatal samples from mice injected with vehicle, haloperidol, or haloperidol + lumateperone is shown in **Figure 2B**

Figure 2. Reduction in Haloperidol-Induced Phosphorylation of TH With Lumateperone Co-Injection

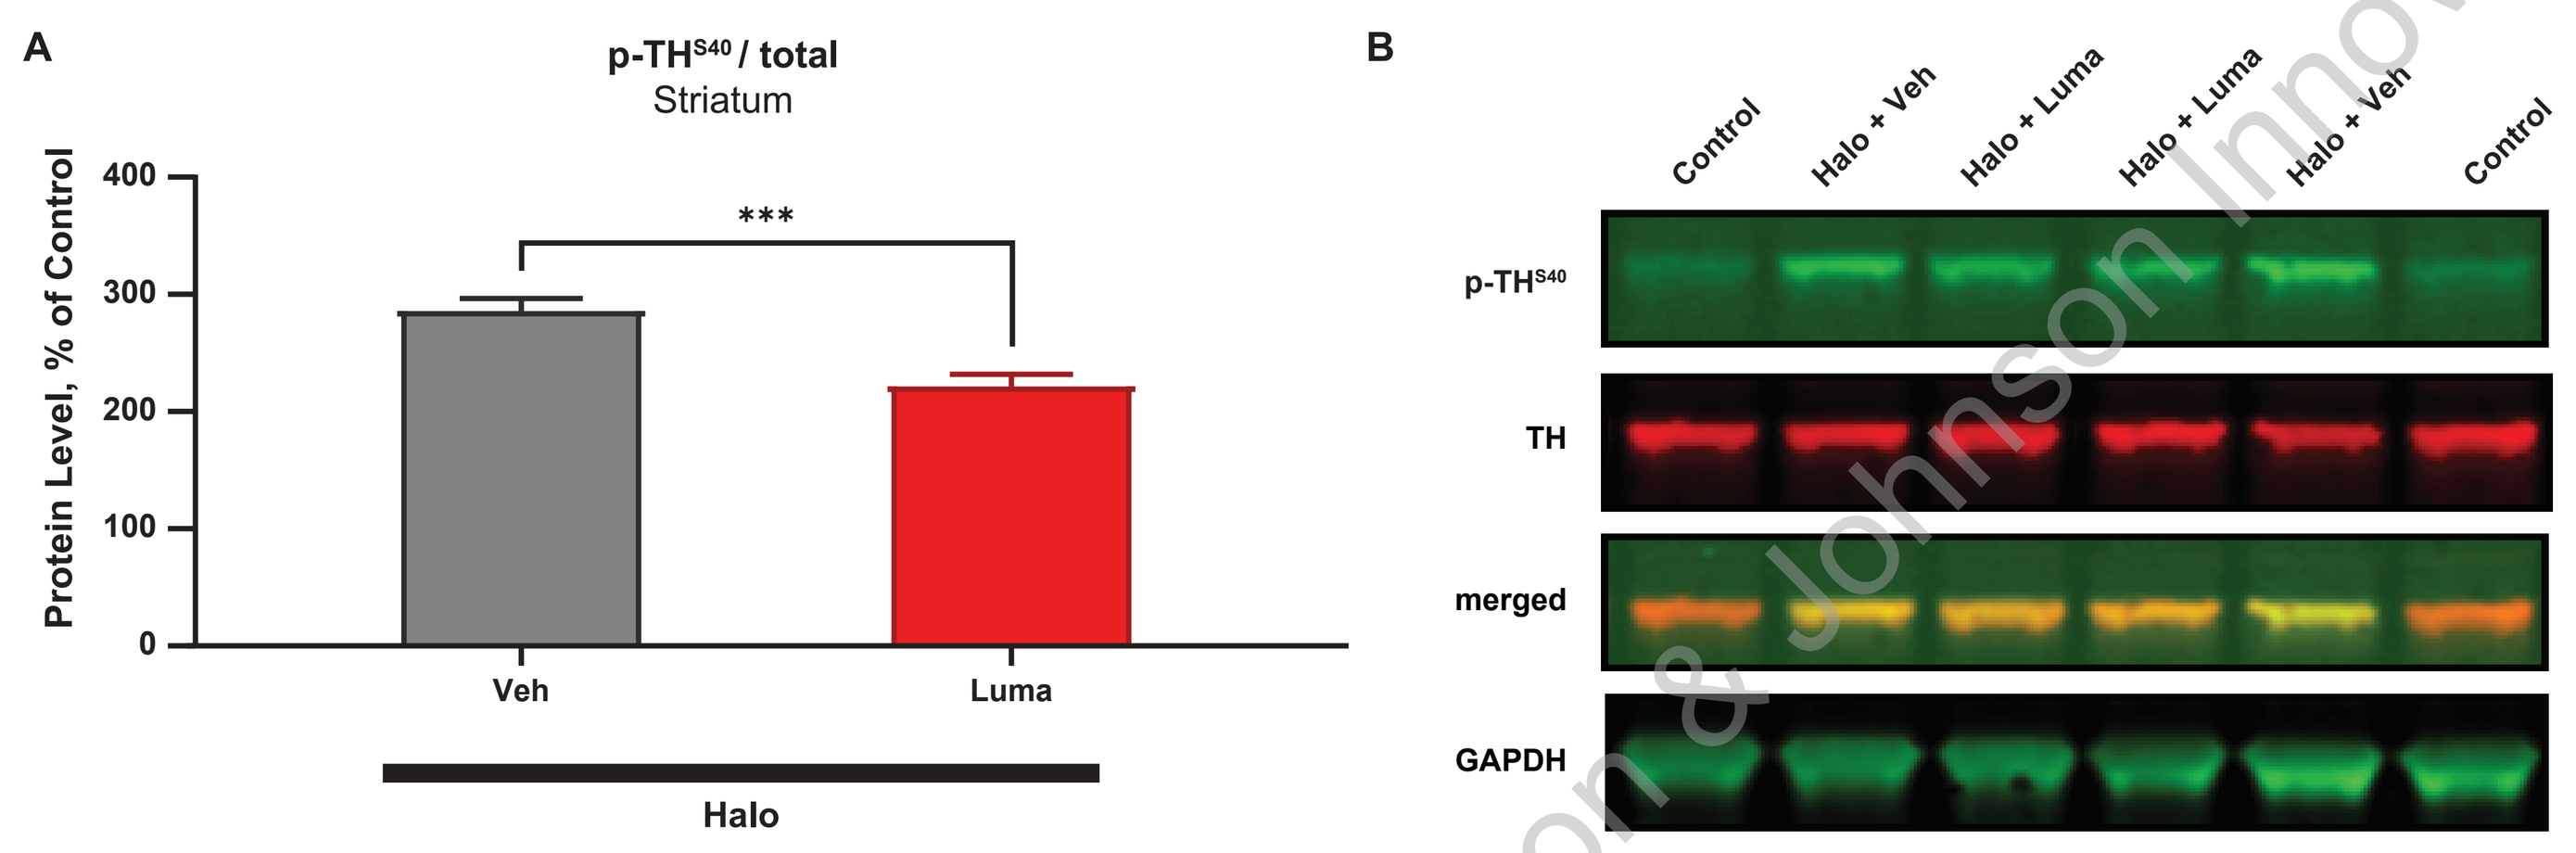
A
p-TH^{S40} / total
Striatum
Protein Level, % of Control

Veh
Luma
Halo
B
Control
Halo + Veh
Halo + Luma
Halo + Luma
Halo + Veh
Control
p-TH^{S40}
TH
merged
GAPDH

P<.01 *P<.001. Control group significantly differed from all other groups (P<.001). Concentrations: Halo 1, mg/kg; Luma, 6 mg/kg.
Halo, haloperidol; Luma, lumateperone; Veh, vehicle.

- The effect of lumateperone in reducing haloperidol-induced p-TH levels in the striatum was replicated by the full D_2 agonist quinpirole, with significant reduction with haloperidol + quinpirole co-injection versus haloperidol alone (P<.05) (**Figure 3A**)
 - Injection of quinpirole alone did not affect basal levels of p-TH (**Figure 3B**)

Figure 3. Co-Administration of a Full D_2 Agonist (Quinpirole) Suppressed Haloperidol-Induced TH Phosphorylation

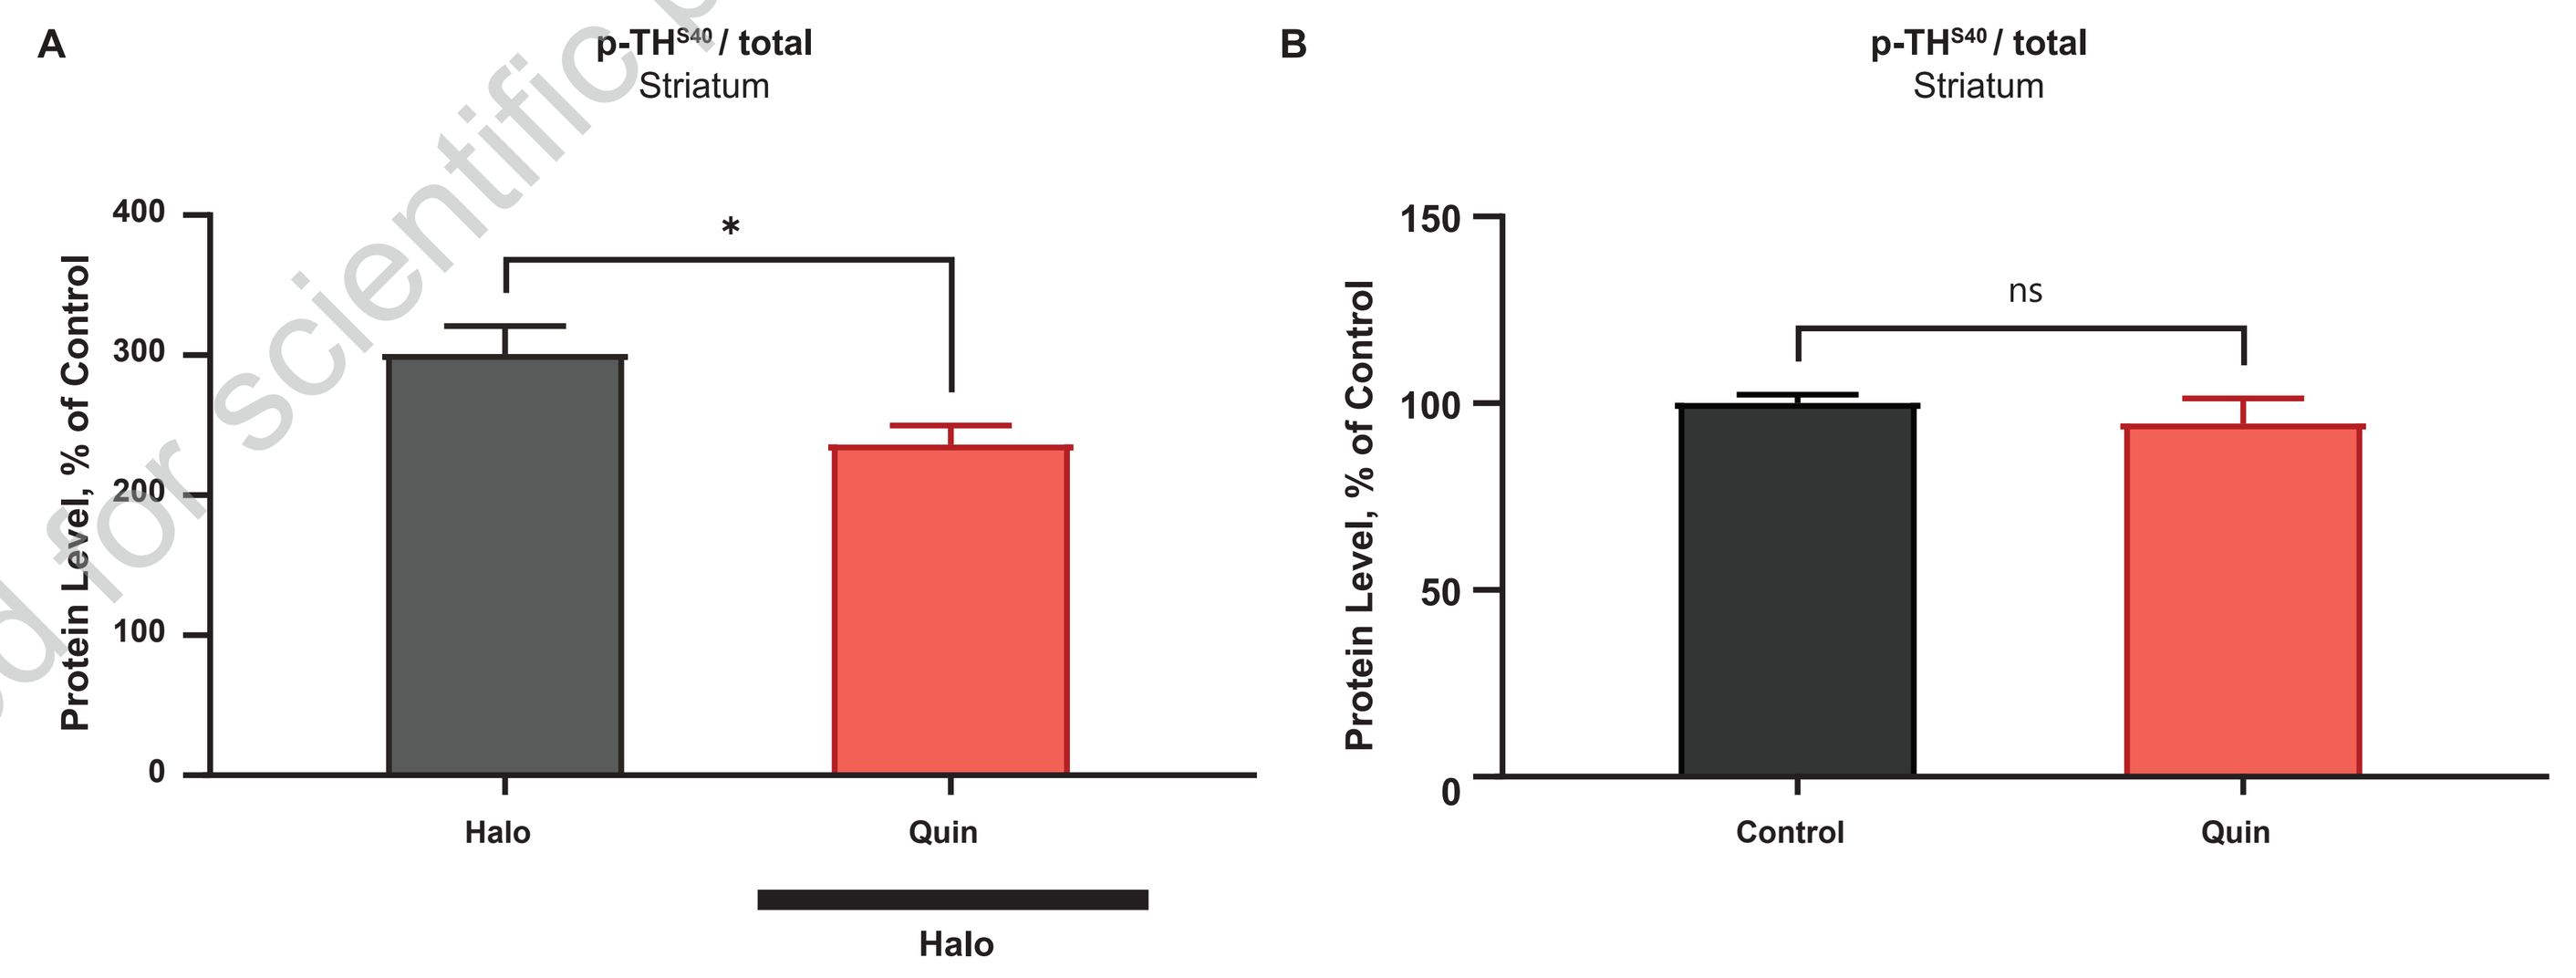
A
p-TH^{S40} / total
Striatum
Protein Level, % of Control
*
Halo
Quin
Halo
B
p-TH^{S40} / total
Striatum
Protein Level, % of Control
ns
Control
Quin

*P<.05. Concentrations: Halo, 1 mg/kg; Quin, 2 mg/kg (Panel A) or 1 mg/kg (Panel B).
Halo, haloperidol; ns, not significant; Quin, quinpirole.

- In this in vivo model, the partial D_2 agonists aripiprazole, brexpiprazole, and cariprazine mimicked the effect of lumateperone and the full agonist quinpirole in reducing haloperidol-induced TH phosphorylation, with cariprazine reaching statistically significant reduction (P<.0001) (**Figure 4**)

Figure 4. Co-Administration of Partial D_2 Agonists Suppressed Haloperidol-Induced TH Phosphorylation

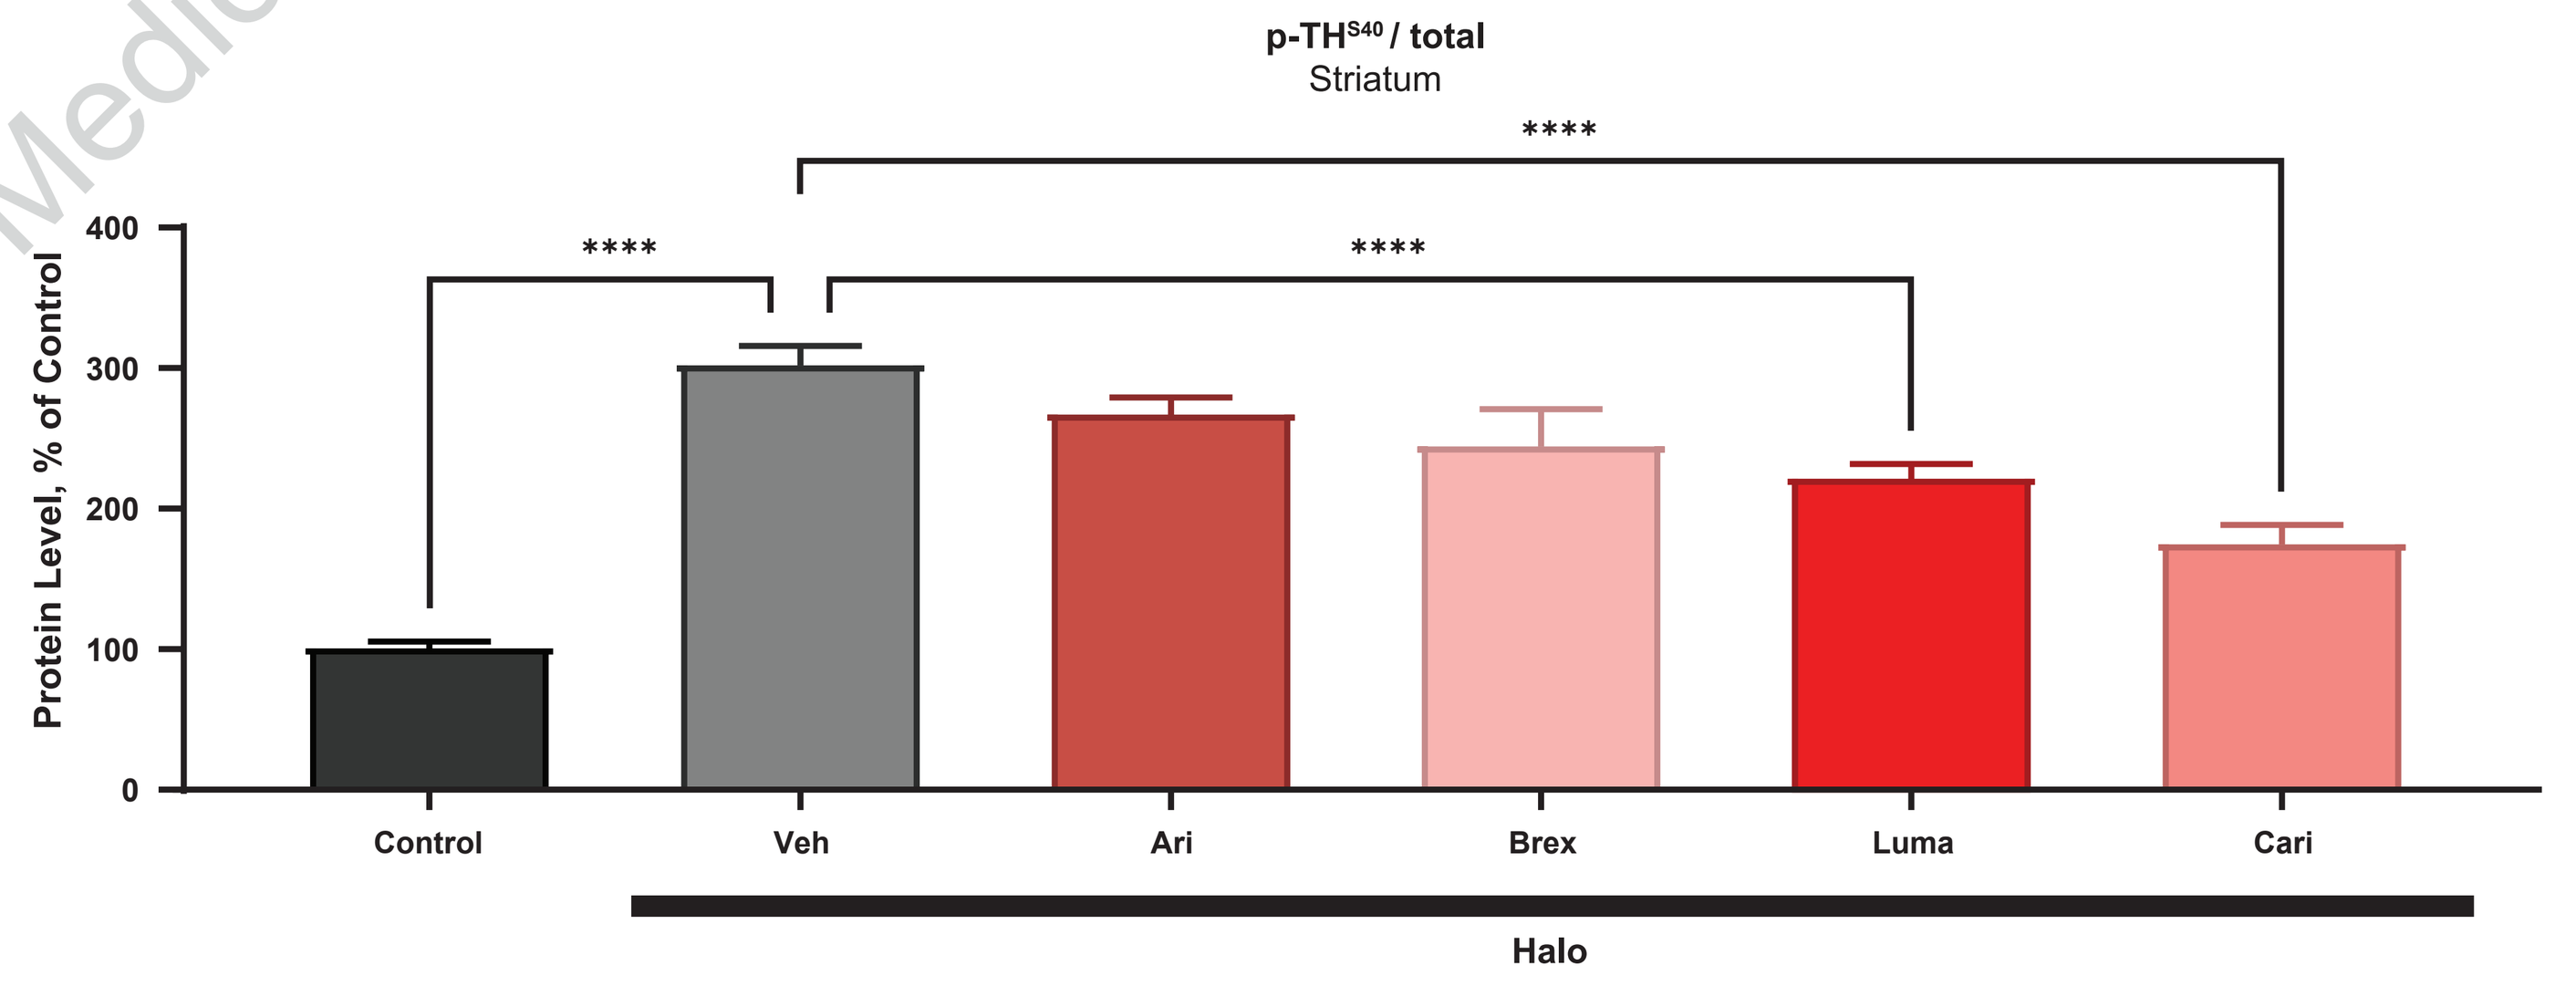
p-TH^{S40} / total
Striatum
Protein Level, % of Control

Control
Veh
Ari
Brex
Luma
Cari
Halo

****P<.0001. Concentrations: Ari, 3 mg/kg; Brex, 10 mg/kg; Cari, 3 mg/kg; Halo, 1 mg/kg; Luma, 6 mg/kg.
Ari, aripiprazole; Brex, brexpiprazole; Cari, cariprazine; Halo, haloperidol; Luma, lumateperone; Veh, vehicle.

- Lurasidone and quetiapine, which are antipsychotics with D_2 antagonist (but not agonist) activity, alone elevated basal phosphorylated TH (**Figure 5A,B**) without suppressing haloperidol-induced phosphorylated TH (**Figure 5C,D**)

Figure 5. Co-Administration of D_2 Antagonists (Lurasidone and Quetiapine) Increased Basal p-TH Levels But Did Not Suppress Haloperidol-Induced p-TH

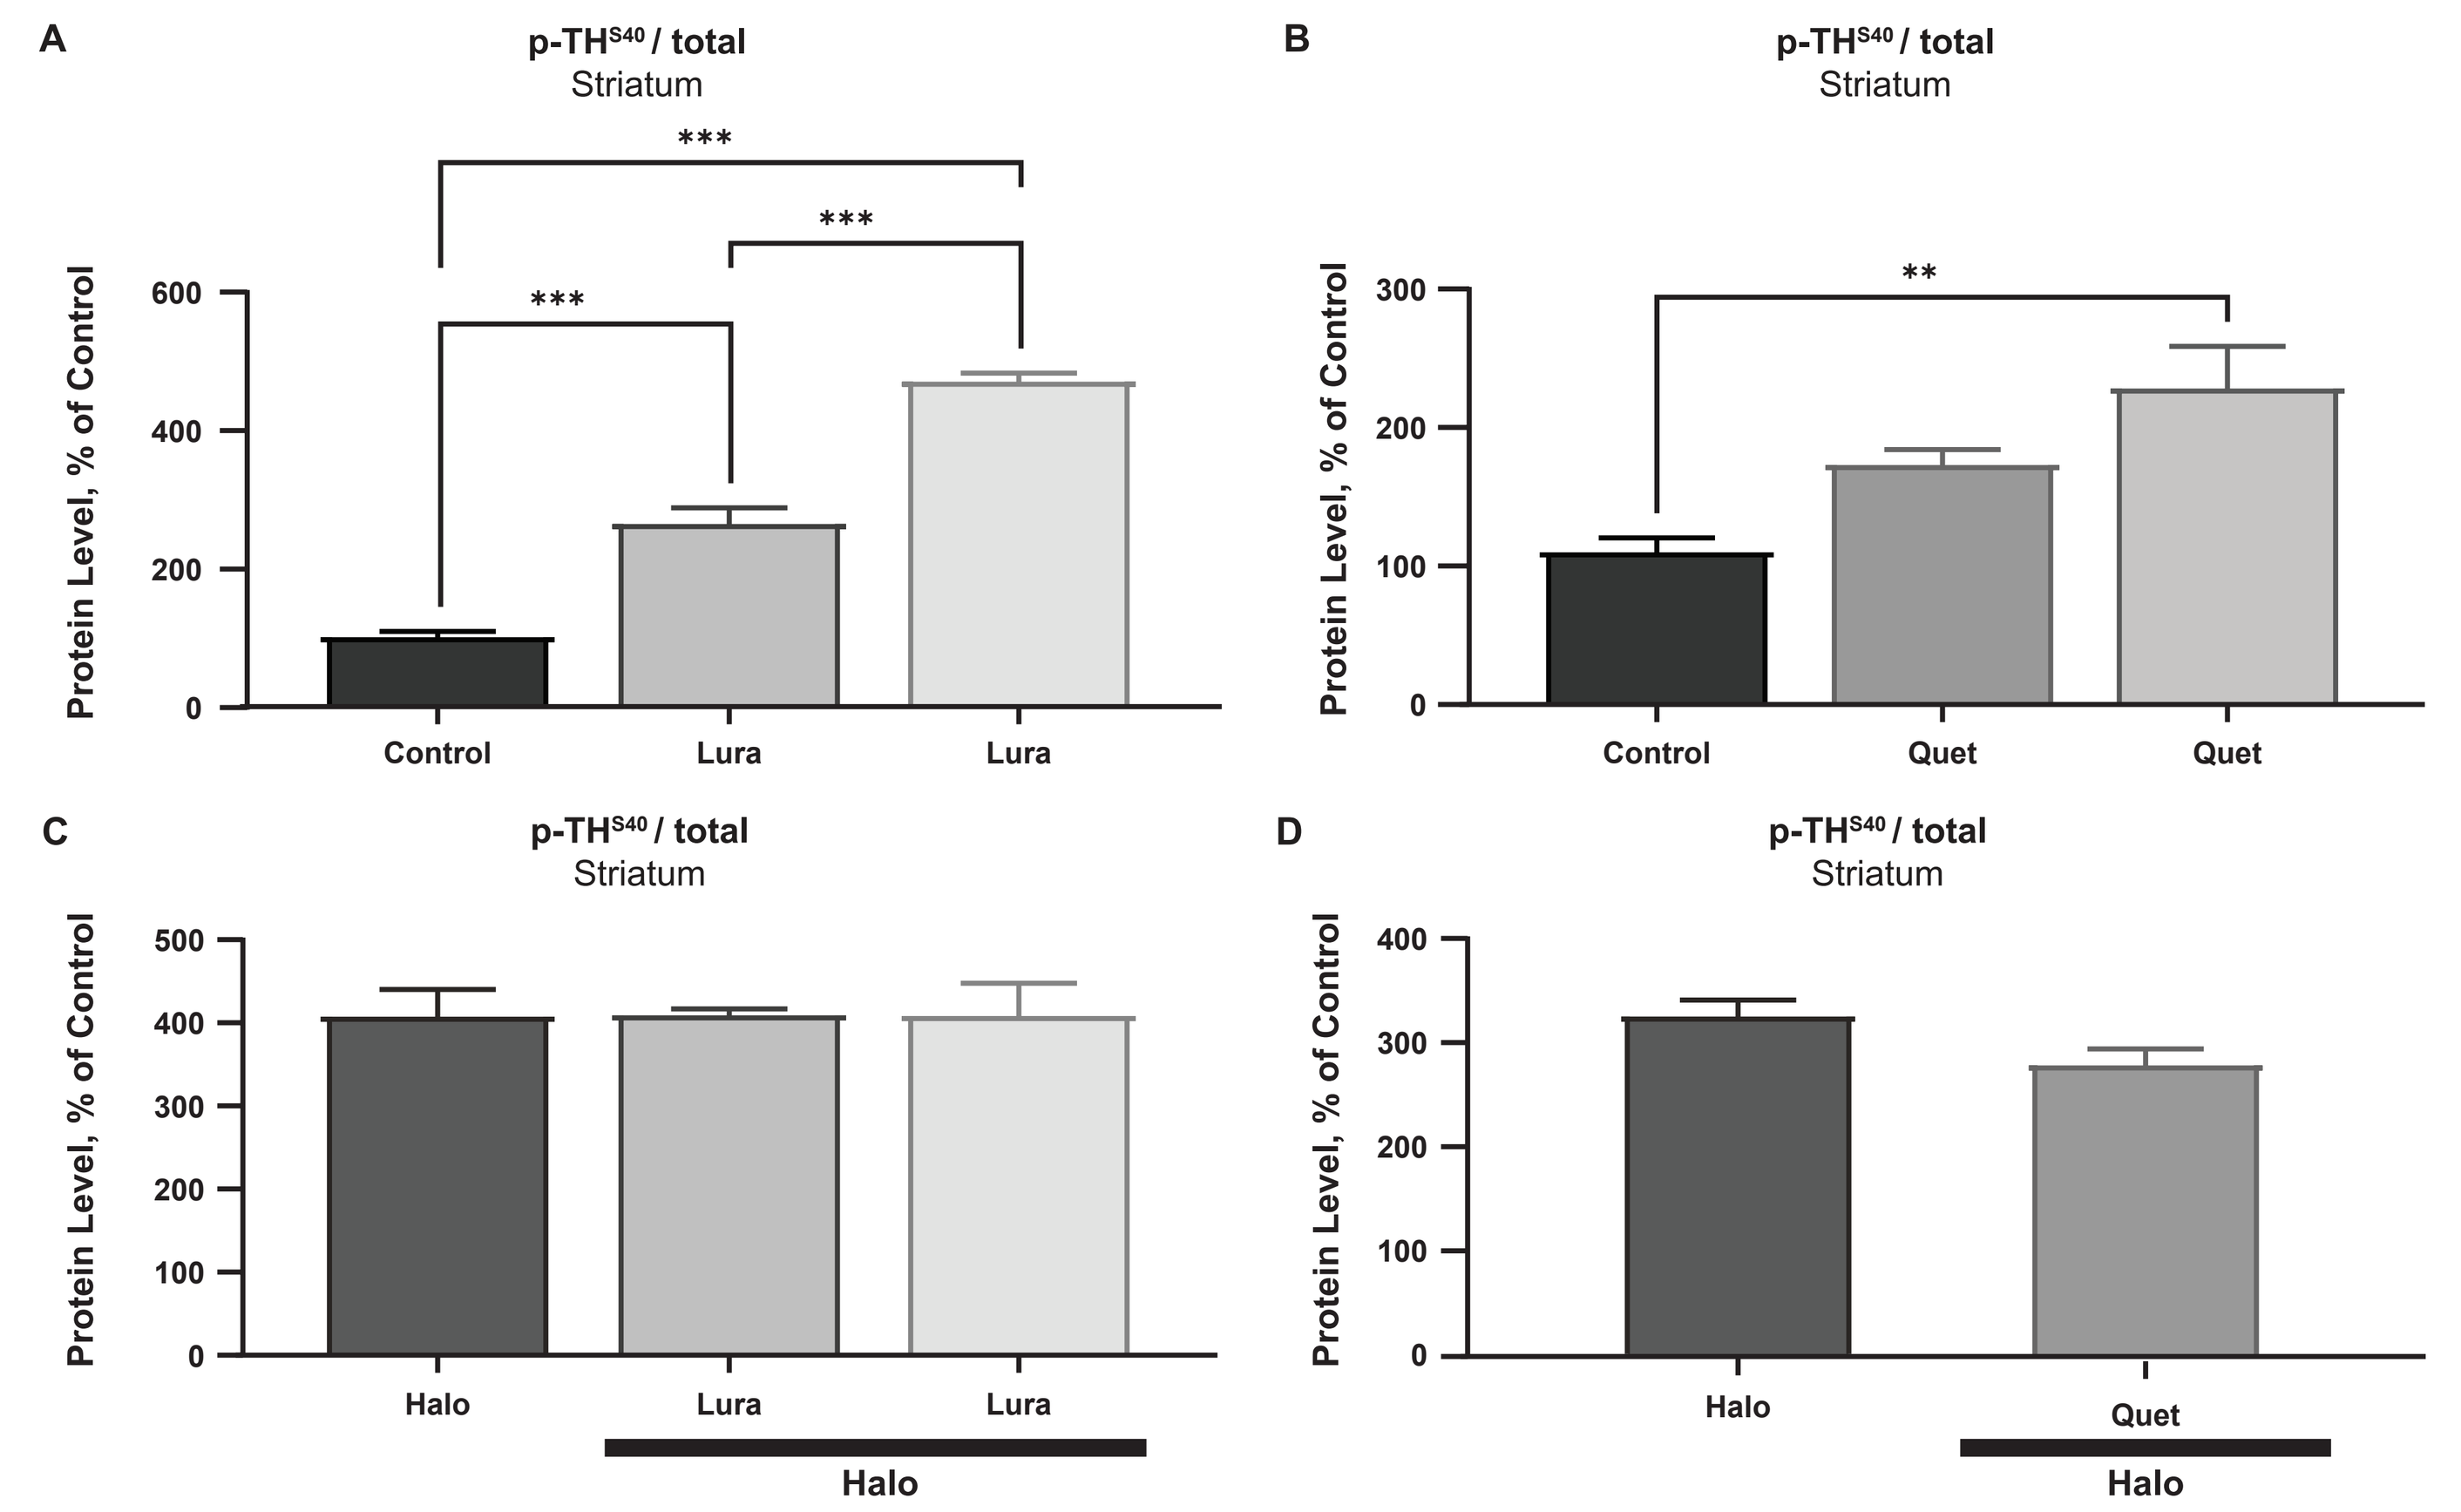
A
p-TH^{S40} / total
Striatum
Protein Level, % of Control

Control
Lura
Lura
B
p-TH^{S40} / total
Striatum
Protein Level, % of Control
**
Control
Quet
Quet
C
p-TH^{S40} / total
Striatum
Protein Level, % of Control
Halo
Lura
Lura
Halo
D
p-TH^{S40} / total
Striatum
Protein Level, % of Control
Halo
Quet
Halo

P<.01 *P<.001. Concentrations: Halo, 1 mg/kg; Lura, 1 mg/kg (darker grey) or 3 mg/kg (lighter grey); Quet, 5 mg/kg (darker grey) or 10 mg/kg (lighter grey).
Halo, haloperidol; Luma, lumateperone; Lura, lurasidone; Quet, quetiapine; Veh, vehicle.

Conclusions

- Lumateperone demonstrated in vivo suppression of haloperidol-induced increases in the phosphorylation of TH at S40, as expressed locally in striatal terminals
- Lumateperone's suppression of haloperidol-induced p-TH occurred in a manner consistent with the actions of full/partial dopamine D_2 receptor agonists (cariprazine, quinpirole, aripiprazole, and brexpiprazole) at presynaptic dopamine D_2 receptors
- In contrast, D_2 antagonists increased p-TH (haloperidol, lurasidone, quetiapine) and did not suppress haloperidol effects (lurasidone, quetiapine)
- The results suggest that lumateperone acts as a dopamine D_2 receptor presynaptic partial agonist and does not act as a dopamine D_2 receptor presynaptic antagonist when measured in vivo
- The unique mechanism of action of lumateperone on the dopaminergic system may contribute to the low risk of EPS and akathisia, and overall favorable safety profile, observed with lumateperone in clinical trials[10]

Acknowledgments
Medical writing support was provided by Keri Ramessar, PhD, of Nucleus Global, an Inizio company, funded by Intra-Cellular Therapies, a Johnson & Johnson company.

Disclosures
M Chen, GL Snyder, and S Dutheil are full-time employees of Johnson & Johnson.

Funding
This study was funded by Intra-Cellular Therapies, a Johnson & Johnson company.

Previously presented at the Society of Biological Psychiatry (SOBP) Annual Meeting, April 30-May 2, 2026, New York, NY, USA.

Neuropsychiatry

Scan the QR code

The QR code is intended to provide scientific information for individual reference, and the information should not be altered or reproduced in any way.

Email questions to
Maxine Chen at MChen158@ITS.JNJ.com

Presented at Psych Congress 2026,
September 15-19, 2026, New Orleans, LA, USA

References:
1. Horacek J, et al. *CNS Drugs.* 2006;20(5):389-409; 2. Ishibashi T, et al. *J Pharmacol Exp Ther.* 2010;334(1):171-181; 3. Cookson J, Pimm J. *BJPsych Advances.* 2023;29:145-150; 4. Vanover KE, et al. *Neuropsychopharmacol.* 2019;44(3):598-605; 5. Titulaer J, et al. *Eur Neuropsychopharmacol.* 2022;62:22-35; 6. Caplyta (lumateperone). Prescribing information. Intra-Cellular Therapies, Inc; 2026; 7. Snyder GL, et al. *Psychopharmacology.* 2015;232(3):605-621; 8. Dunkley PR, et al. *J Neurochem.* 2019;149:706-728; 9. Håkansson K, et al. *Eur J Neurosci.* 2004;20(4):1108-1112; 10. Kane JM, et al. *Int Clin Psychopharmacol.* 2021;36(5):244-250; 11. Leys C, et al. *J Exp Soc Psychol.* 2013;49(4):764-766.

This material is distributed for scientific purposes by Johnson & Johnson Innovative Medicine and is not for promotional use.