111

Adjunctive Lumateperone 42 mg Treatment in Major Depressive Disorder: Efficacy Across Patient-Reported Depression Symptoms

Rakesh Jain, MD[1]; Suresh Durgam, MD[2]; Willie R. Earley, MD[3]; Susan G. Kozauer, MD[2]; Changzheng Chen, PhD[3]

[1]Department of Psychiatry, Texas Tech University School of Medicine – Permian Basin, Midland, TX, USA; [2]Former employee of Intra-Cellular Therapies, a Johnson & Johnson company, Bedminster, NJ, USA; [3]Intra-Cellular Therapies, a Johnson & Johnson company, Bedminster, NJ, USA

This material is distributed for scientific purposes by Johnson & Johnson Innovative Medicine and is not for promotional use.

Background

- Major depressive disorder (MDD) is a complex mental illness characterized by heterogeneous and highly burdensome symptoms[1,2]
- In research and clinical practice, it is important to assess the patient's perspective of symptom burden through patient-reported outcome tools, such as The Quick Inventory of Depressive Symptomatology-Self Report-16 item (QIDS-SR-16) scale[3,4]
 - This validated tool allows patients to rate the severity of their depression symptoms over multiple symptom domains[4,5]
- Many patients, particularly those with anxious distress, experience ongoing symptoms despite treatment with antidepressant therapy (ADT), highlighting the need for treatments that reduce symptom severity across a range of symptom domains in MDD[2,6]
- Lumateperone is an atypical antipsychotic indicated for the treatment of MDD in adults as adjunct to ADT; the treatment of schizophrenia in adults; and the treatment of depressive episodes associated with bipolar I or II disorder (bipolar depression) in adults, as monotherapy and as adjunctive therapy with lithium or valproate[7]
 - Lumateperone is a potent serotonin 5-HT_{2A} receptor antagonist, a dopamine D_2 receptor presynaptic partial agonist and postsynaptic antagonist, a D1 receptor-dependent indirect modulator of glutamatergic AMPA and NMDA currents, and a serotonin reuptake inhibitor[8]
 - This novel mechanism of action with multimodal effects may confer robust efficacy with improved tolerability compared with current treatment options
- In a positive, Phase 3, randomized, double-blind, placebo-controlled trial (Study 501 [NCT04985942]), adjunctive lumateperone 42 mg demonstrated significant improvement in the Montgomery-Åsberg Depression Rating Scale (MADRS) Total score versus adjunctive placebo at Day 43 and was generally well tolerated in patients with MDD with inadequate ADT response[9]
 - Additionally, patients treated with lumateperone + ADT vs placebo + ADT reported significant improvement in QIDS-SR-16 Total score at Day 43
- In this post hoc analysis of Study 501, the efficacy of lumateperone 42 mg + ADT vs placebo + ADT was evaluated across the QIDS-SR-16 symptom domains in patients with MDD with inadequate ADT response in the overall population and within clinical subgroups

Methods

- Eligible adults (aged 18-65 years) met DSM-5 criteria for MDD with inadequate response to 1-2 ADTs in the current depressive episode (<50% improvement on the Antidepressant Treatment Response Questionnaire) and had MADRS Total score ≥24, Clinical Global Impression-Severity score ≥4, and QIDS-SR-16 Total score ≥14
- Patients were randomized (1:1) to receive 6-week lumateperone 42 mg or placebo adjunctive to ADT
- A prespecified analysis measured change from baseline to Day 43 in QIDS-SR-16 Total score using an analysis of covariance (ANCOVA) in the intent-to-treat (ITT) population
- This post hoc analysis measured change from baseline to end of treatment (EOT) of the 9 mutually exclusive QIDS-SR-16 domain scores, using an ANCOVA in the overall ITT population and within patient subgroups according to baseline disease characteristics (DSM-5–defined anxious distress [yes vs no] and disease severity [MADRS Total score <32 vs ≥32])
- The 16 QIDS-SR-16 items correspond with the 9 DSM-5 symptom domains used as the diagnostic criteria for MDD[2,3] (**Figure 1**)
 - For each QIDS-SR-16 item, symptom severity was reported on a scale from 0 to 3, with higher scores indicating more severe depression symptoms[3]
 - For the 3 symptom domains comprising >1 QIDS-SR-16 item (i.e., sleep disturbance, appetite/weight change, and psychomotor agitation/retardation), the domain score was defined as the highest observed score across all included items[3]

Figure 1. DSM-5 MDD Symptom Criterion Domains and Corresponding QIDS-SR-16 Items[2,3]

DSM-5 MDD Symptom Criterion Domains	QIDS-SR-16 Items
Sleep disturbances	Highest score of: 1. Falling asleep 2. Sleep during the night 3. Waking up too early 4. Sleeping too much
Sad mood	5. Feeling sad
Decrease/increase in appetite/weight	Highest score of: 6. Decreased appetite 7. Increased appetite 8. Decreased weight 9. Increased weight
Concentration	10. Concentration/decision making
Self-criticism	11. View of myself
Suicidal ideation	12. Thoughts of death or suicide
Interest	13. General interest
Energy/fatigue	14. Energy level
Psychomotor agitation/retardation	Higher score of: 15. Feeling slowed down 16. Feeling restless

DSM-5, *Diagnostic and Statistical Manual of Mental Disorders, Fifth Edition*; MDD, major depressive disorder; QIDS-SR-16, Quick Inventory of Depressive Symptomatology-Self Report-16 item.

Results

Overall Population

- Of 484 patients in the ITT population (lumateperone + ADT, n=241; placebo + ADT, n=243), 93.4% (n=452) completed treatment
- Baseline characteristics were similar between treatment arms[9]
 - Overall, the mean patient age was 45 years; most patients were female (65.7%) and White (76.7%)
- Lumateperone + ADT significantly improved QIDS-SR-16 Total score vs placebo + ADT from baseline to Day 43 (least squares mean difference vs placebo [LSMD], −2.4; effect size [ES], −0.50; *P*<.0001)[9]
- Baseline QIDS-SR-16 symptom domain scores were balanced between treatment groups, with patients reporting the greatest symptom severity in the domains of sleep disturbance, sad mood, concentration, interest, and energy/fatigue (**Figure 2**)

Figure 2. QIDS-SR-16 Symptom Domain Scores at Baseline (ITT Population)

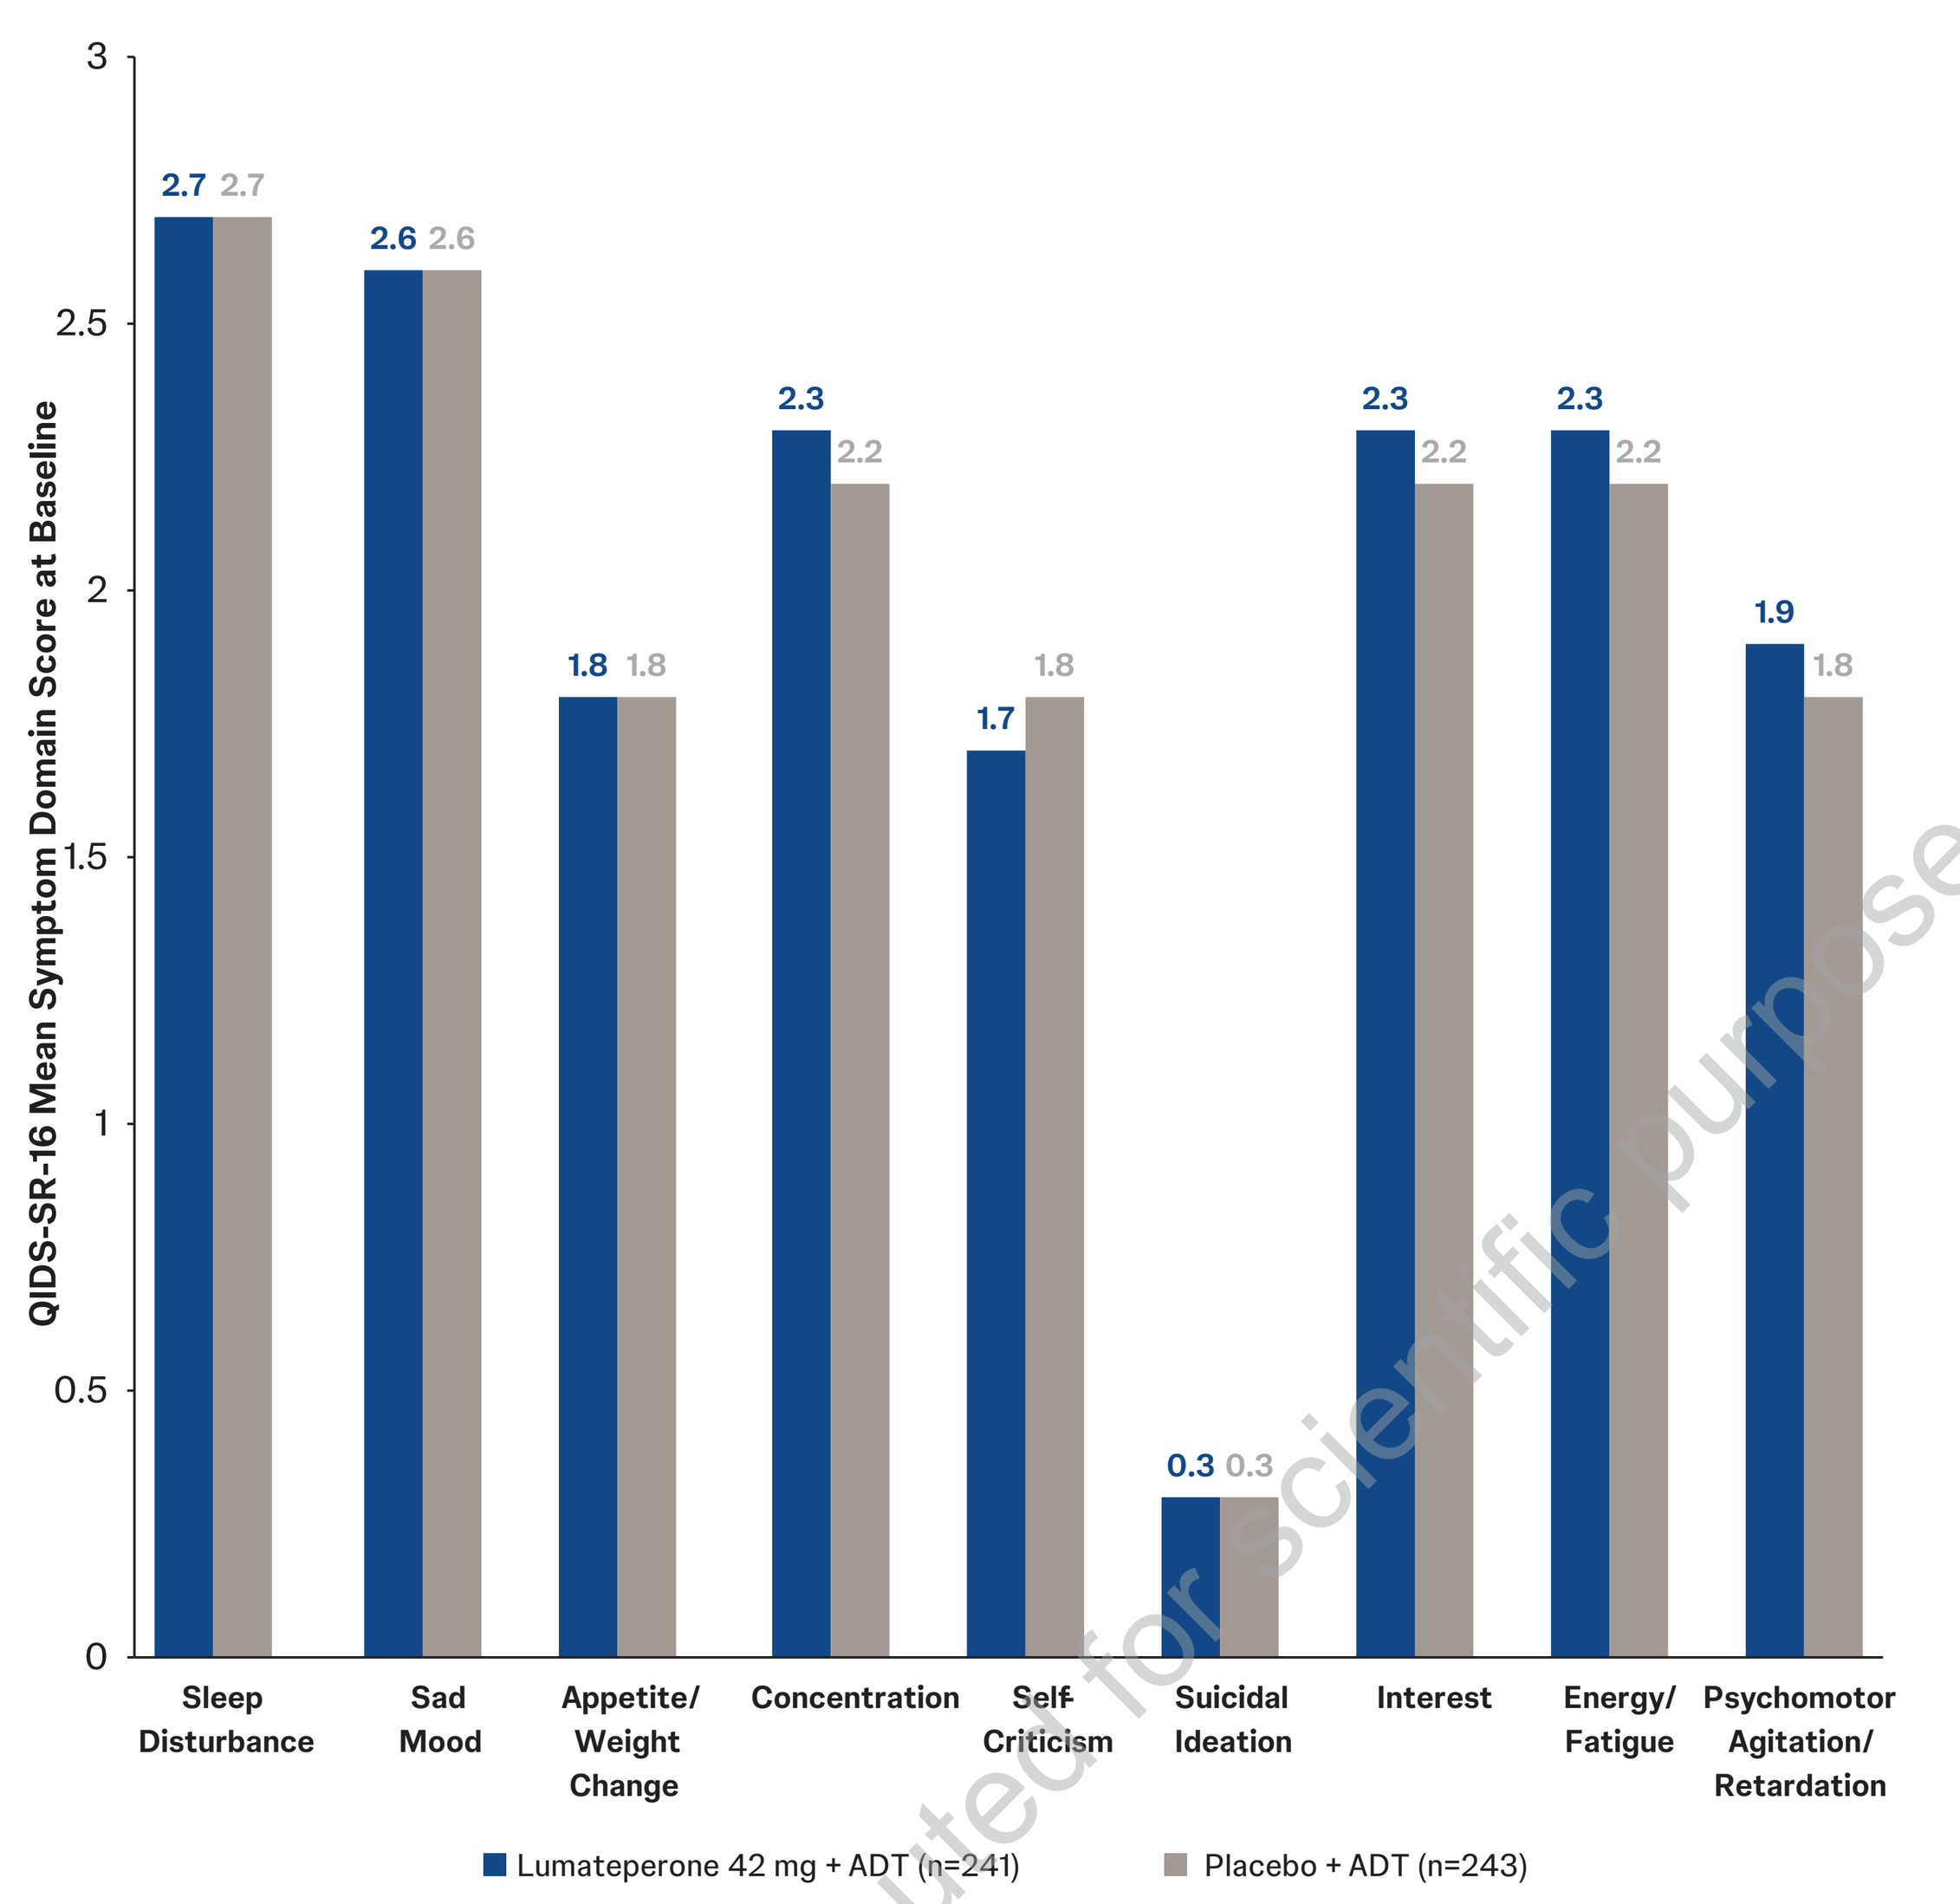
QIDS-SR-16 Mean Symptom Domain Score at Baseline
3
2.5
2
1.5
1
0.5
0
2.7 2.7
2.6 2.6
1.8 1.8
2.3 2.2
1.7 1.8
0.3 0.3
2.3 2.2
2.3 2.2
1.9 1.8
Sleep Disturbance
Sad Mood
Appetite/ Weight Change
Concentration
Self-Criticism
Suicidal Ideation
Interest
Energy/ Fatigue
Psychomotor Agitation/ Retardation
Lumateperone 42 mg + ADT (n=241)
Placebo + ADT (n=243)

ADT, antidepressant therapy; ITT, intent-to-treat; QIDS-SR-16, Quick Inventory of Depressive Symptomatology-Self Report-16 item.

- From baseline to EOT, 8 of the 9 QIDS-SR-16 symptom domain scores significantly improved with lumateperone + ADT vs placebo + ADT (**Figure 3**)
 - Significance was not reached for the change in symptom severity for the domain of self-criticism (*P*=.065)

Figure 3. Change From Baseline to EOT in QIDS-SR-16 Symptom Domain Scores (ITT Population)

	Sleep Disturbance	Sad Mood	Appetite/ Weight Change	Concentration	Self-Criticism	Suicidal Ideation	Interest	Energy/ Fatigue	Psychomotor Agitation/ Retardation
Effect Size	−0.55	−0.44	−0.22	−0.28	−0.17	−0.19	−0.29	−0.29	−0.30
LSMD	−0.47	−0.36	−0.19	−0.22	−0.15	−0.07	−0.25	−0.24	−0.23
Mean Baseline Score	2.7 2.7	2.6 2.6	1.8 1.8	2.3 2.2	1.7 1.8	0.3 0.3	2.3 2.2	2.3 2.2	1.9 1.8

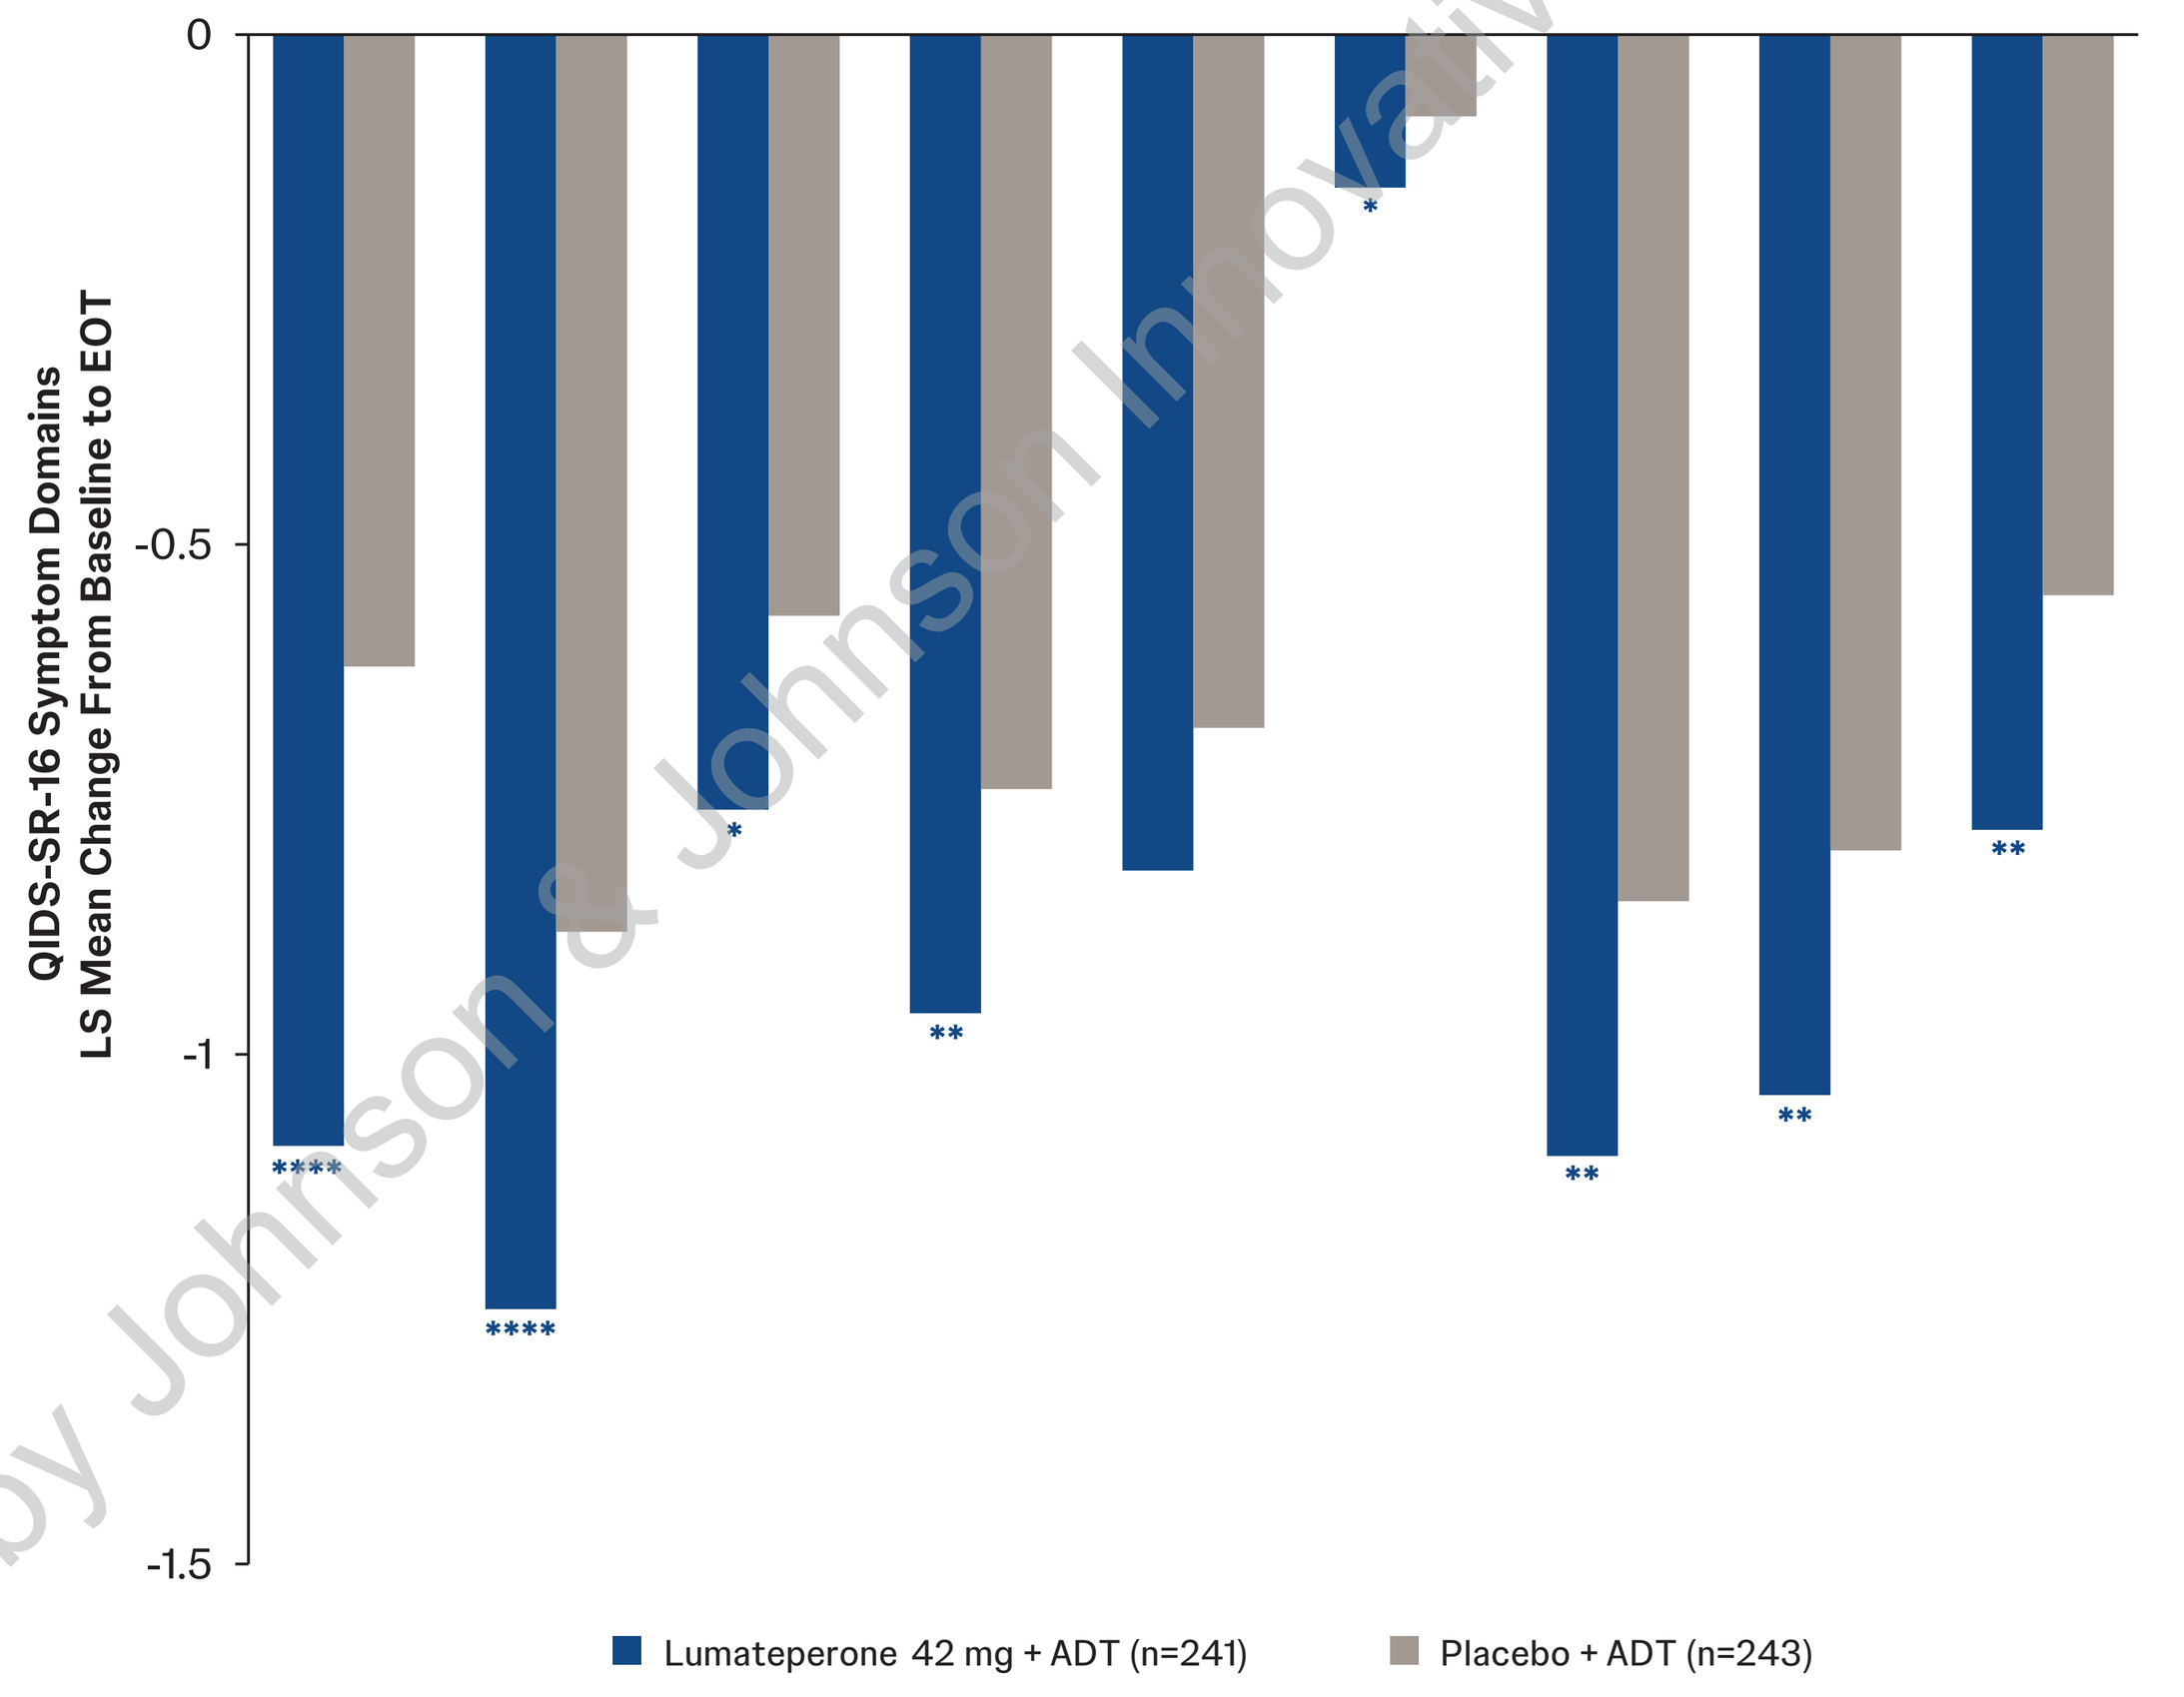
QIDS-SR-16 Symptom Domains LS Mean Change From Baseline to EOT
0
-0.5
-1
-1.5

*
**
*
**
**
**
Lumateperone 42 mg + ADT (n=241)
Placebo + ADT (n=243)

*P<.05 **P<.01 ****P<.0001. ANCOVA in the ITT population.
ADT, antidepressant therapy; ANCOVA, analysis of covariance; EOT, end of treatment; ITT, intent-to-treat; LS, least squares; LSMD, least squares mean difference; QIDS-SR-16, Quick Inventory of Depressive Symptomatology-Self Report-16 item.

Subgroup Analyses

- At EOT, lumateperone + ADT significantly improved 8 of 9 QIDS-SR-16 symptom domains in patients with DSM-5–defined anxious distress (**Figure 4A**)
 - Numerical improvement in symptom domain scores occurred in those without anxious distress, although results were only significant for sleep disturbance (**Figure 4B**)
- In patients with baseline MADRS Total score <32, lumateperone + ADT significantly improved 6 QIDS-SR-16 symptoms domain scores (sleep disturbance, sad mood, concentration, suicidal ideation, interest, and psychomotor agitation/retardation; [*P*<.05]) from baseline to EOT
 - Patients with baseline MADRS Total score ≥32 had significant improvement (*P*<.05) with lumateperone + ADT at EOT in the QIDS-SR-16 symptom domain scores for sleep disturbance, sad mood, concentration, energy/fatigue, and psychomotor agitation/retardation

Figure 4. Change From Baseline to EOT in QIDS-SR-16 Symptom Domain Scores in Patient Subgroups per Presence/Absence of Anxious Distress at Baseline (ITT Population)

A. Patients With Anxious Distress

	Sleep Disturbance	Sad Mood	Appetite/ Weight Change	Concentration	Self-Criticism	Suicidal Ideation	Interest	Energy/ Fatigue	Psychomotor Agitation/ Retardation
Effect Size	−0.91	−0.69	−0.39	−0.47	−0.30	−0.21	−0.45	−0.43	−0.47
LSMD	−0.76	−0.58	−0.33	−0.37	−0.26	−0.08	−0.39	−0.36	−0.35
Mean Baseline Score	2.8 2.8	2.7 2.6	1.8 1.8	2.4 2.3	1.9 2.0	0.4 0.3	2.4 2.3	2.3 2.2	2.0 1.9

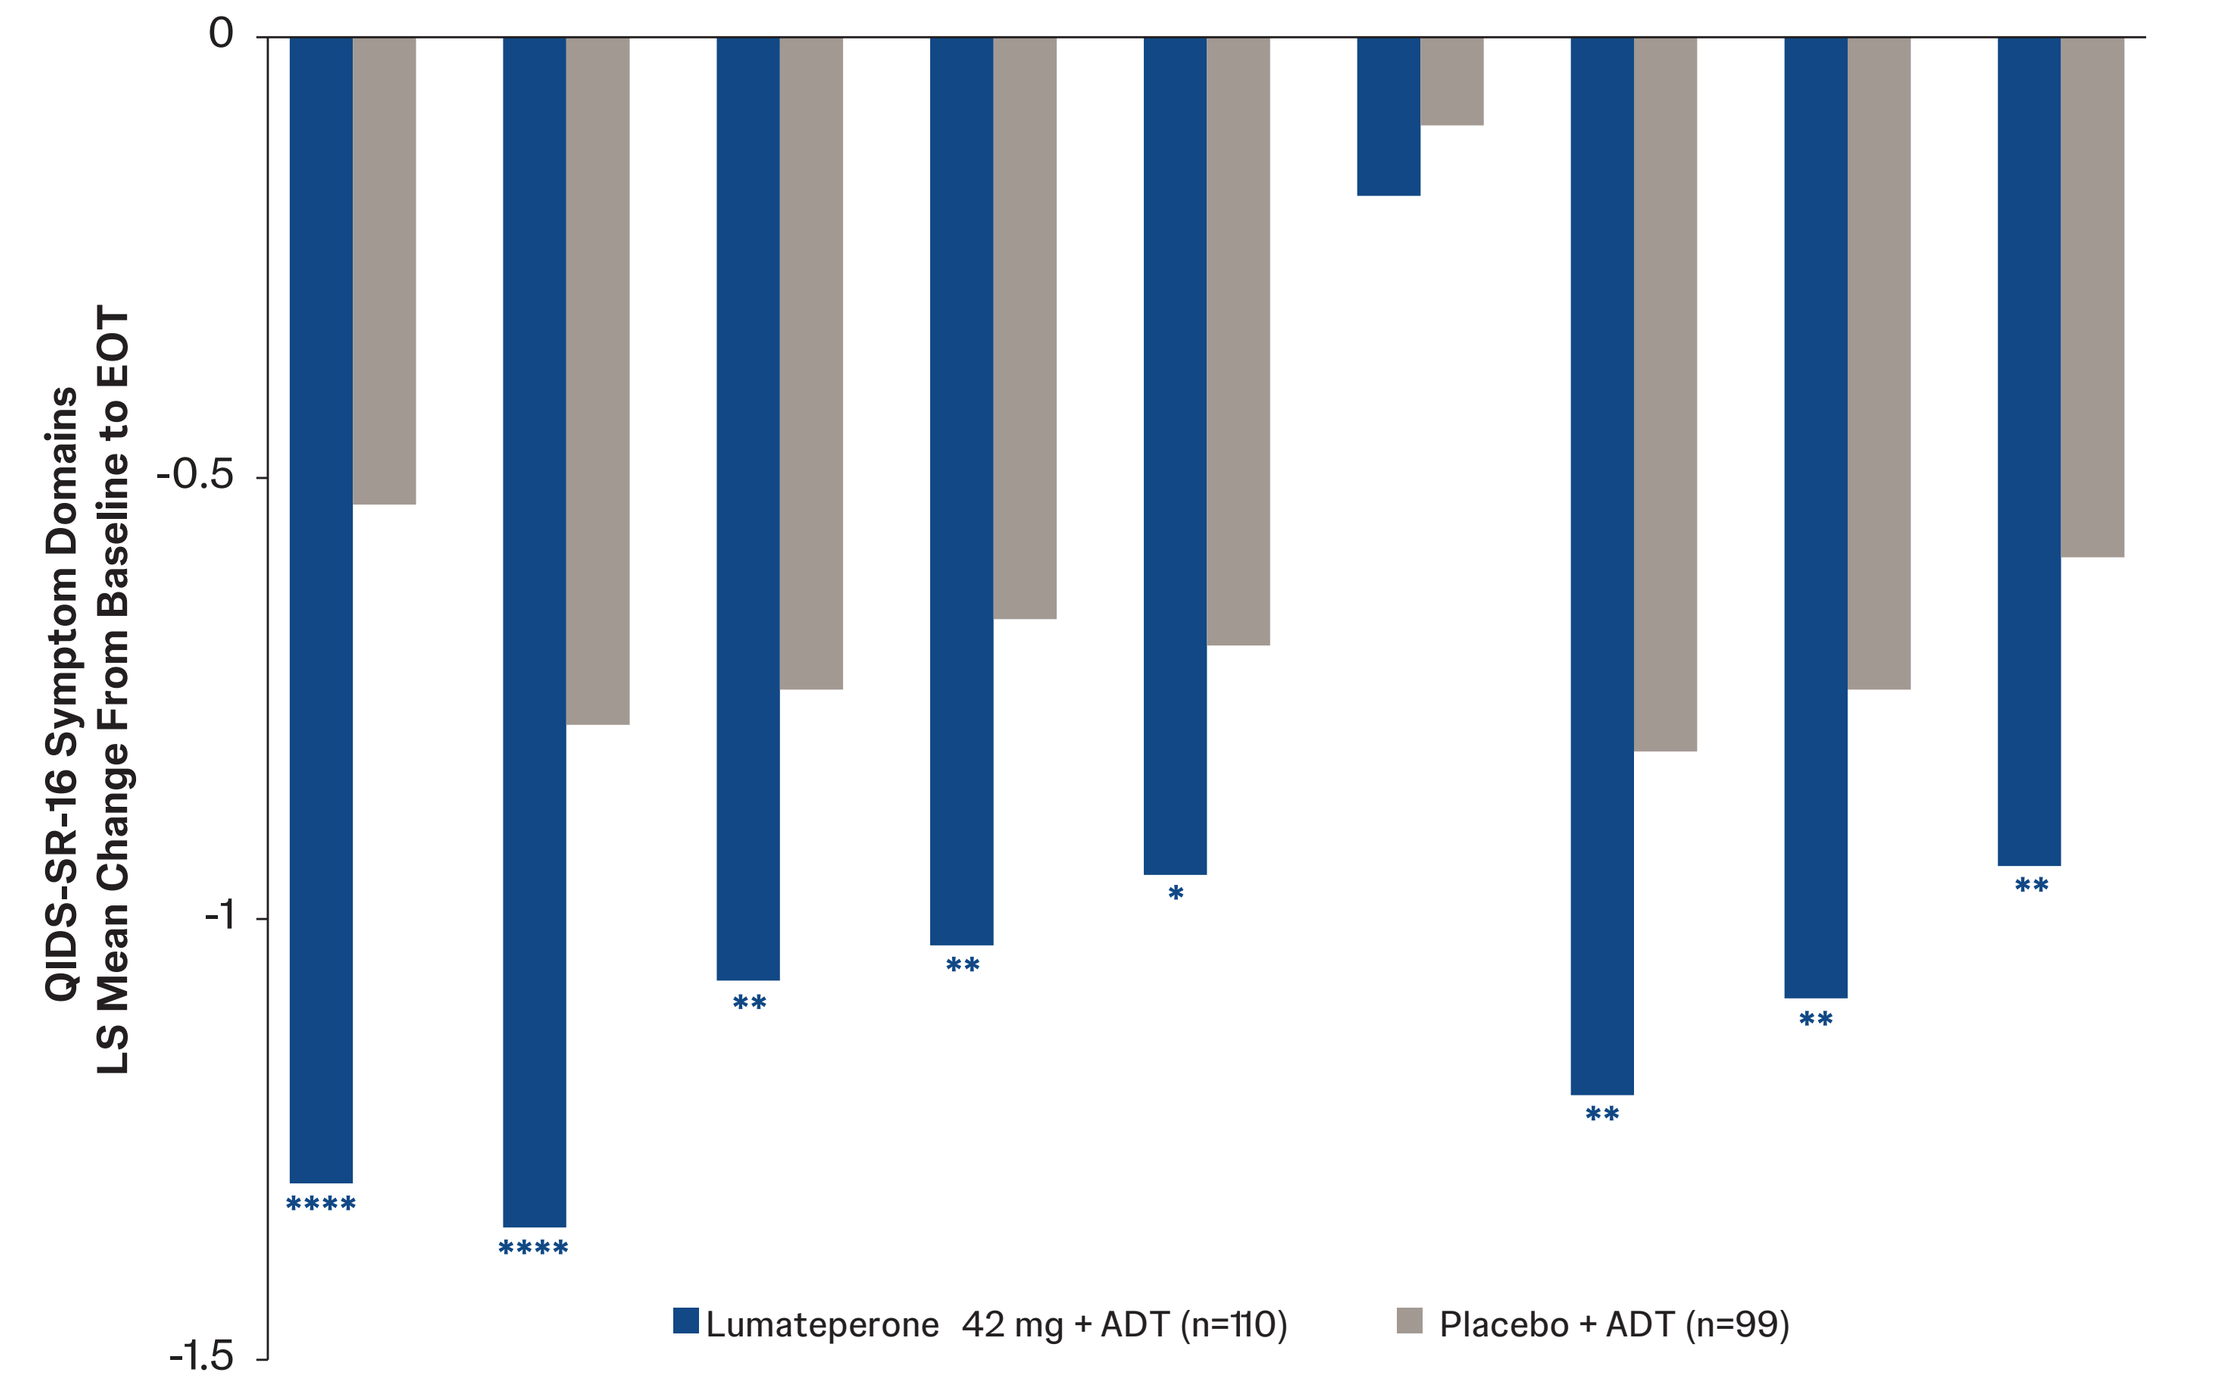
QIDS-SR-16 Symptom Domains LS Mean Change From Baseline to EOT
0
-0.5
-1
-1.5

**
**
*
**
**
**
Lumateperone 42 mg + ADT (n=110)
Placebo + ADT (n=99)

B. Patients Without Anxious Distress

	Sleep Disturbance	Sad Mood	Appetite/ Weight Change	Concentration	Self-Criticism	Suicidal Ideation	Interest	Energy/ Fatigue	Psychomotor Agitation/ Retardation
Effect Size	−0.28	−0.24	−0.05	−0.13	−0.06	−0.17	−0.16	−0.19	−0.17
LSMD	−0.24	−0.20	−0.05	−0.11	−0.05	−0.06	−0.14	−0.16	−0.13
Mean Baseline Score	2.7 2.7	2.6 2.6	1.8 1.7	2.2 2.2	1.6 1.7	0.2 0.3	2.3 2.2	2.2 2.2	1.8 1.8

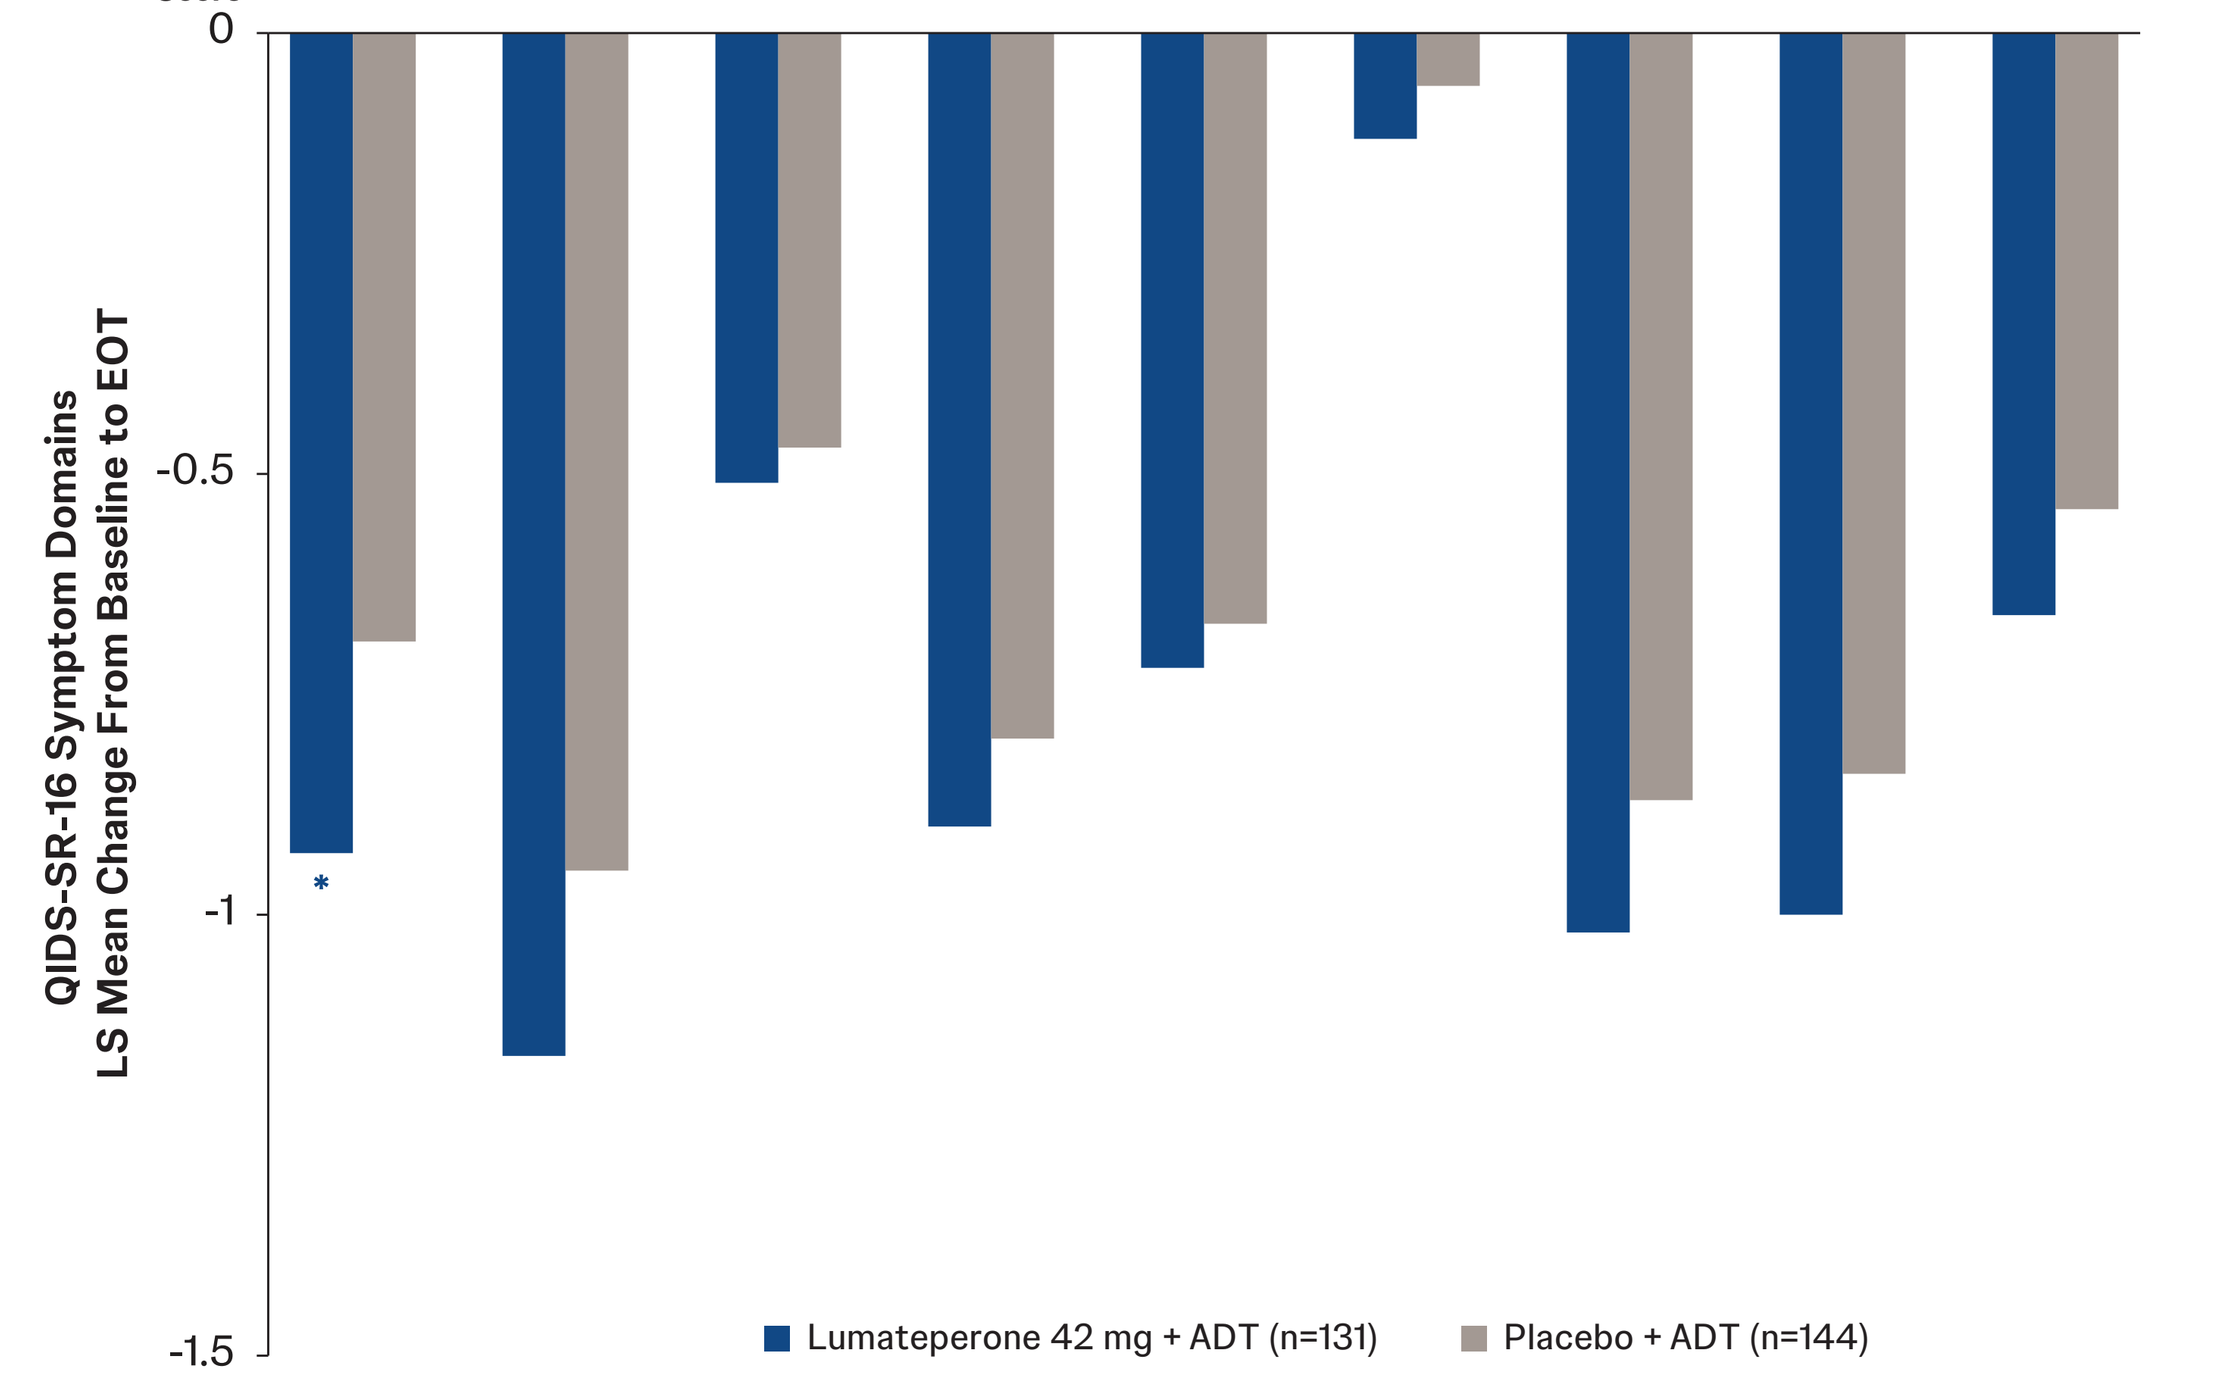
QIDS-SR-16 Symptom Domains LS Mean Change From Baseline to EOT
0
-0.5
-1
-1.5
*
Lumateperone 42 mg + ADT (n=131)
Placebo + ADT (n=144)

*P<.05 **P<.01 ****P<.0001. ANCOVA in the ITT population.
ADT, antidepressant therapy; ANCOVA, analysis of covariance; EOT, end of treatment; ITT, intent-to-treat; LS, least squares; LSMD, least squares mean difference; QIDS-SR-16, Quick Inventory of Depressive Symptomatology-Self Report-16 item.

Conclusions

- In Study 501, lumateperone 42 mg + ADT showed significant improvements over placebo + ADT in depression symptom severity across 8 of 9 QIDS-SR-16 symptom domains
- Significant improvements occurred with lumateperone 42 mg + ADT in sleep disturbance, which was the most prominent symptom domain at baseline
- Lumateperone 42 mg + ADT significantly improved QIDS-SR-16 symptom domains in patients with anxious distress and across groups of baseline disease severity
- These findings support the efficacy of adjunctive lumateperone in patients with MDD with inadequate ADT response across a range of symptom domains in MDD

Acknowledgments

The authors thank all study investigators, research staff, and patients for their participation. Medical writing support was provided by Joy Steenkamp, PharmD, of Nucleus Global, an Inizio company, funded by Intra-Cellular Therapies, a Johnson & Johnson company.

Disclosures

R Jain has served as a consultant to Addrenex, Allergan (now AbbVie), Avanir, Janssen, Lilly, Lundbeck, Merck, Neos Therapeutics, Neurocrine Biosciences, Otsuka, Pamlab, Pfizer, Shionogi, Shire, Sunovion, Supernus, Takeda, and Teva; paid speaker for Addrenex, Alkermes, Allergan (now AbbVie), Lilly, Lundbeck, Merck, Neos Therapeutics, Otsuka, Pamlab, Pfizer, Rhodes, Shionogi, Shire, Sunovion, Takeda, and Tris Pharmaceuticals; received research support from Allergan (now AbbVie), AstraZeneca, Lilly, Lundbeck, Otsuka, Pfizer, Shire, and Takeda; and served on advisory board for Addrenex, Alkermes, Avanir, Forum, Janssen, Lilly, Lundbeck, Merck, Neos Therapeutics, Neurocrine Biosciences, Otsuka, Pamlab, Pfizer, Shionogi, Shire, Sunovion, Supernus, Takeda, and Teva.

WR Earley and C Chen are full-time employees of Intra-Cellular Therapies, a Johnson & Johnson company.

S Durgam and SG Kozauer are former employees of Intra-Cellular Therapies, a Johnson & Johnson company.

Funding

This study was funded by Intra-Cellular Therapies, a Johnson & Johnson company.

Previously presented at the American Society of Clinical Psychopharmacology (ASCP) Annual Meeting, May 26-29, 2026, Miami, FL, USA.

Neuropsychiatry

Scan the QR code

The QR code is intended to provide scientific information for individual reference, and the information should not be altered or reproduced in any way.

Email questions to Willie R. Earley at wearley@its.jnj.com

Presented at Psych Congress 2026, September 15-19, 2026, New Orleans, LA, USA

References:
1. Su YA, Si T. *Gen Psychiatry*. 2022;35:e100724; 2. Malhi GS, Mann JJ. *Lancet*. 2018;392:2299-2312; 3. Rush AJ, et al. *Biol Psychiatry*. 2006;59:493-501; 4. Palmer EOC, et al. *Neuropsychiatr Dis Treat*. 2024;20:671-687; 5. Rush AJ, et al. *Biol Psychiatry*. 2003;54:573-583; 6. Brown TM, et al. *J Depress Anxiety*. 2016;5:3; 7. Caplyta (lumateperone). Prescribing information. Intra-Cellular Therapies, Inc; 2026; 8. Titulaer J, et al. *Eur Neuropsychopharmacol*. 2022;62:22-35; 9. Durgam S, et al. *J Clin Psychiatry*. 2025;86:25m15848.