Adjunctive Lumateperone 42 mg Treatment in Major Depressive Disorder: A Pooled Analysis of Efficacy Across Patient-Reported Depression Symptoms

Michael E. Thase, MD[1]; Ibrahim Turkoz, PhD[2]; Brian Curcio, MS[3]; Kristin R. Dorsey, PharmD[2]; Zhizheng Zhang, MS[2]; Manish K. Jha, MD[4]

[1]Perelman School of Medicine, University of Pennsylvania, Philadelphia, PA, USA; [2]Johnson & Johnson, Titusville, NJ, USA; [3]ICON plc. Philadelphia, PA, USA; [4]Department of Psychiatry, University of Texas Southwestern Medical Center, Dallas, TX, USA

99

This material is distributed for scientific purposes by Johnson & Johnson Innovative Medicine and is not for promotional use.

Background

- Major depressive disorder (MDD) is a complex mental illness characterized by heterogeneous and highly burdensome symptoms[1,2]
- Many patients experience ongoing symptoms despite treatment with antidepressant therapy (ADT), highlighting the need for treatments that reduce symptom severity across a range of symptom domains in MDD[1,2]
- Assessing symptom severity using patient-reported outcome measures is important in both clinical research and the management of MDD[3,4]
 - The Quick Inventory of Depressive Symptomatology-Self Report 16-item (QIDS-SR-16) is a validated patient-reported outcome measure that assesses overall depressive symptom severity and individual symptom domains[4,5]
- Lumateperone is FDA approved for the treatment of MDD in adults as an adjunctive therapy to ADT; treatment of schizophrenia in adults; and treatment of depressive episodes associated with bipolar I or II disorder (bipolar depression) in adults, as monotherapy and as an adjunctive therapy with lithium or valproate[6]
 - Lumateperone is a mechanistically novel, multimodal antipsychotic that modulates serotonergic, dopaminergic, and, indirectly, glutamatergic neurotransmission[6,7]
- In 2 Phase 3, randomized, double-blind, placebo-controlled studies (Study 501, NCT04985942; Study 502, NCT05061706), adjunctive lumateperone 42 mg provided superior efficacy versus adjunctive placebo, with meaningful and significant improvements in depressive symptoms (Montgomery-Åsberg Depression Rating Scale [MADRS] Total score) and disease severity (Clinical Global Impression Scale – Severity [CGI-S] score) and was generally well tolerated in patients with MDD with inadequate ADT response[8,9]
 - Additionally, lumateperone + ADT significantly improved QIDS-SR-16 Total score at Day 43 versus placebo + ADT
- This pooled analysis of Studies 501 and 502 assessed the efficacy of adjunctive lumateperone 42 mg across individual depressive symptom domains in patients with MDD, as measured by QIDS-SR-16 scores collected at baseline and Day 43/end of treatment (EOT)

Methods

- Eligible adults (aged 18-65 years) in Studies 501 and 502 met *Diagnostic and Statistical Manual of Mental Disorders, Fifth Edition* (DSM-5) criteria for MDD with inadequate response to 1 or 2 ADT courses (<50% improvement on the Antidepressant Treatment Response Questionnaire) in the current depressive episode and had MADRS Total score ≥24, Clinical Global Impression-Severity score ≥4, and QIDS-SR-16 Total score ≥14[8,9]
- Patients were randomized (1:1) to receive 6 weeks of oral lumateperone 42 mg or placebo per day adjunctive to ADT[8,9]

Methods (cont)

- A prespecified analysis measured change from baseline to Day 43 in QIDS-SR-16 Total score using an analysis of covariance (ANCOVA) in the intent-to-treat (ITT) population
- The 16 QIDS-SR-16 items correspond with the 9 DSM-5 symptom domains used as the diagnostic criteria for MDD[2,3] (**Figure 1**)
 - For each QIDS-SR-16 item, symptom severity was reported on a scale from 0 to 3, with higher scores indicating more severe depression symptoms[3]
 - For the 3 symptom domains comprising >1 QIDS-SR-16 item (ie, sleep disturbance, appetite/weight change, and psychomotor agitation/retardation), the domain score was defined as the highest observed score across all included items[3]
- This post hoc analysis assessed the following changes in QIDS-SR-16 scores from baseline to EOT pooled from both studies using ANCOVA:
 - Changes from baseline to EOT in the QIDS-SR-16 Total score, individual domain scores, and interest-activity factor score
 - The anhedonia-related interest-activity factor score was defined as the sum of QIDS-SR-16 items 10 (concentration/decision-making), 13 (general interest), and 14 (energy level)
 - QIDS-SR-16 Total scores according to response (≥50% improvement in Total score from baseline to EOT) and remission (Total score ≤5 at EOT) status
 - The proportion of patients achieving ≥1- and ≥2-point improvement within each QIDS-SR-16 domain at EOT
 - Domain-level analyses were not adjusted for multiplicity; therefore, all reported *P* values are nominal

Figure 1. DSM-5 MDD Symptom Criterion Domains and Corresponding QIDS-SR-16 Items[4,5]

DSM-5 MDD Symptom Criterion Domains	QIDS-SR-16 Items
Sleep disturbances	Highest score of: 1. Falling asleep 2. Sleep during the night 3. Waking up too early 4. Sleeping too much
Sad mood	5. Feeling sad
Decrease/increase in appetite/weight	Highest score of: 6. Decreased appetite 7. Increased appetite 8. Decreased weight 9. Increased weight
Concentration	10. Concentration/decision making
Self-criticism	11. View of myself
Suicidal ideation	12. Thoughts of death or suicide
Interest	13. General interest
Energy/fatigue	14. Energy level
Psychomotor agitation/retardation	Highest score of: 15. Feeling slowed down 16. Feeling restless

DSM-5, *Diagnostic and Statistical Manual of Mental Disorders, Fifth Edition*; MDD, major depressive disorder; QIDS-SR-16, Quick Inventory of Depressive Symptomatology-Self Report-16 item.

Results

Patient Population

- Of 950 patients in the modified intent-to-treat (mITT) population (lumateperone + ADT, n=471; placebo + ADT, n=479), 92.7% (n=881) completed treatment
- Baseline characteristics were similar between treatment arms (**Table 1**)
 - The majority of patients were female (67.7%), and 86.0% were White

Table 1. Baseline Demographics and Clinical Characteristics (mITT Population)[10]

	Lumateperone 42 mg + ADT (n=471)	Placebo + ADT (n=479)
Age, mean (range), years	45.2 (18-65)	45.8 (18-65)
Sex, n (%)		
Women	318 (67.5)	325 (67.8)
Men	153 (32.5)	154 (32.2)
Race, n (%)		
White	404 (85.8)	413 (86.2)
Asian	41 (8.7)	35 (7.3)
Black	25 (5.3)	24 (5.0)
Other[a]	1 (0.2)	7 (1.5)
Hispanic or Latino ethnicity, n (%)	50 (10.6)	49 (10.2)
Number of lifetime depressive episodes, mean (range)	3.6 (1-30)	4.0 (1-36)
Lifetime history of treatment failures including the current MDE, n (%)		
1	352 (74.7)	352 (73.5)
2	119 (25.3)	127 (26.5)
QIDS-SR-16 Total score, mean (SD)	18.0 (2.42)	17.8 (2.39)

[a]Other includes Native Hawaiian or other Pacific Islander, multiple, or other.
ADT, antidepressant therapy; MDE, major depressive episode; mITT, modified intent-to-treat; QIDS-SR-16, Quick Inventory of Depressive Symptoms-Self Report 16 Items.

QIDS-SR-16 Total Score/QIDS-SR-16 Response and Remission

- Lumateperone + ADT significantly improved QIDS-SR-16 Total score versus placebo + ADT from baseline to Day 43
- QIDS-SR-16 Total score mean (median) change from baseline was −8.1 (−8.0) with lumateperone + ADT versus −5.7 (−5.0) with placebo +ADT (least squares mean difference versus placebo [LSMD], −2.37; effect size, 0.47; *P*<.001) (**Table 2**)
- A greater proportion of patients achieved QIDS-SR-16 response and remission with lumateperone + ADT versus placebo + ADT (**Table 2**)

Table 2. Change From Baseline to EOT in QIDS-SR-16 Total Score and QIDS-SR-16 Response[a] and Remission[b]

	Lumateperone 42 mg + ADT (n=471)	Placebo + ADT (n=479)
QIDS-SR-16 Total score, mean (SD)		
At baseline	18.0 (2.42)	17.8 (2.39)
At EOT	9.9 (5.23)	12.2 (5.15)
LS mean change from baseline to EOT (SE)	−8.1 (0.23)	−5.7 (0.23)
LSMD (SE)	−2.37 (0.329)	
Effect size	0.47	
P value[c]	<.001	
Response,[a] n/N (%)	221/460 (48.0)	138/470 (29.4)
Odds ratio (95% CI)	2.22 (1.70-2.91)	
P value	<.001[d]	
Remission,[b] n/N (%)	97/460 (21.1)	49/470 (10.4)
Odds ratio (95% CI)	2.30 (1.58-3.33)	
P value	<.001[d]	

[a]Reponse is defined as ≥50% reduction QIDS-SR-16 Total score from baseline to EOT. [b]Remission is defined as QIDS-SR-16 Total score ≤5. [c]ANCOVA in the mITT population. [d]Cochran-Mantel-Haenszel test comparing treatment groups.
ADT, antidepressant therapy; ANCOVA, analysis of covariance; CI, confidence interval; EOT, end of treatment; LS, least squares; LSMD, least squares mean difference; mITT, modified intent-to-treat; QIDS-SR-16, Quick Inventory of Depressive Symptomatology-Self Report-16 item.

QIDS-SR-16 Symptom Domains

- Significant improvements were observed across all 9 QIDS-SR-16 symptom domains with lumateperone + ADT versus placebo + ADT (LSMD, −0.06 to −0.56; effect size, 0.16 to 0.58; all *P*<.05) (**Figure 2**)
 - The greatest improvements were observed for sleep disturbance (LSMD, −0.56; effect size, 0.58, *P*<.001), sad mood (LSMD, −0.37; effect size, 0.43, *P*<.001), and interest (LSMD, −0.28; effect size, 0.31; *P*<.001) (**Figure 2**)

Figure 2. Change From Baseline to EOT in QIDS-SR-16 Symptom Domain Scores

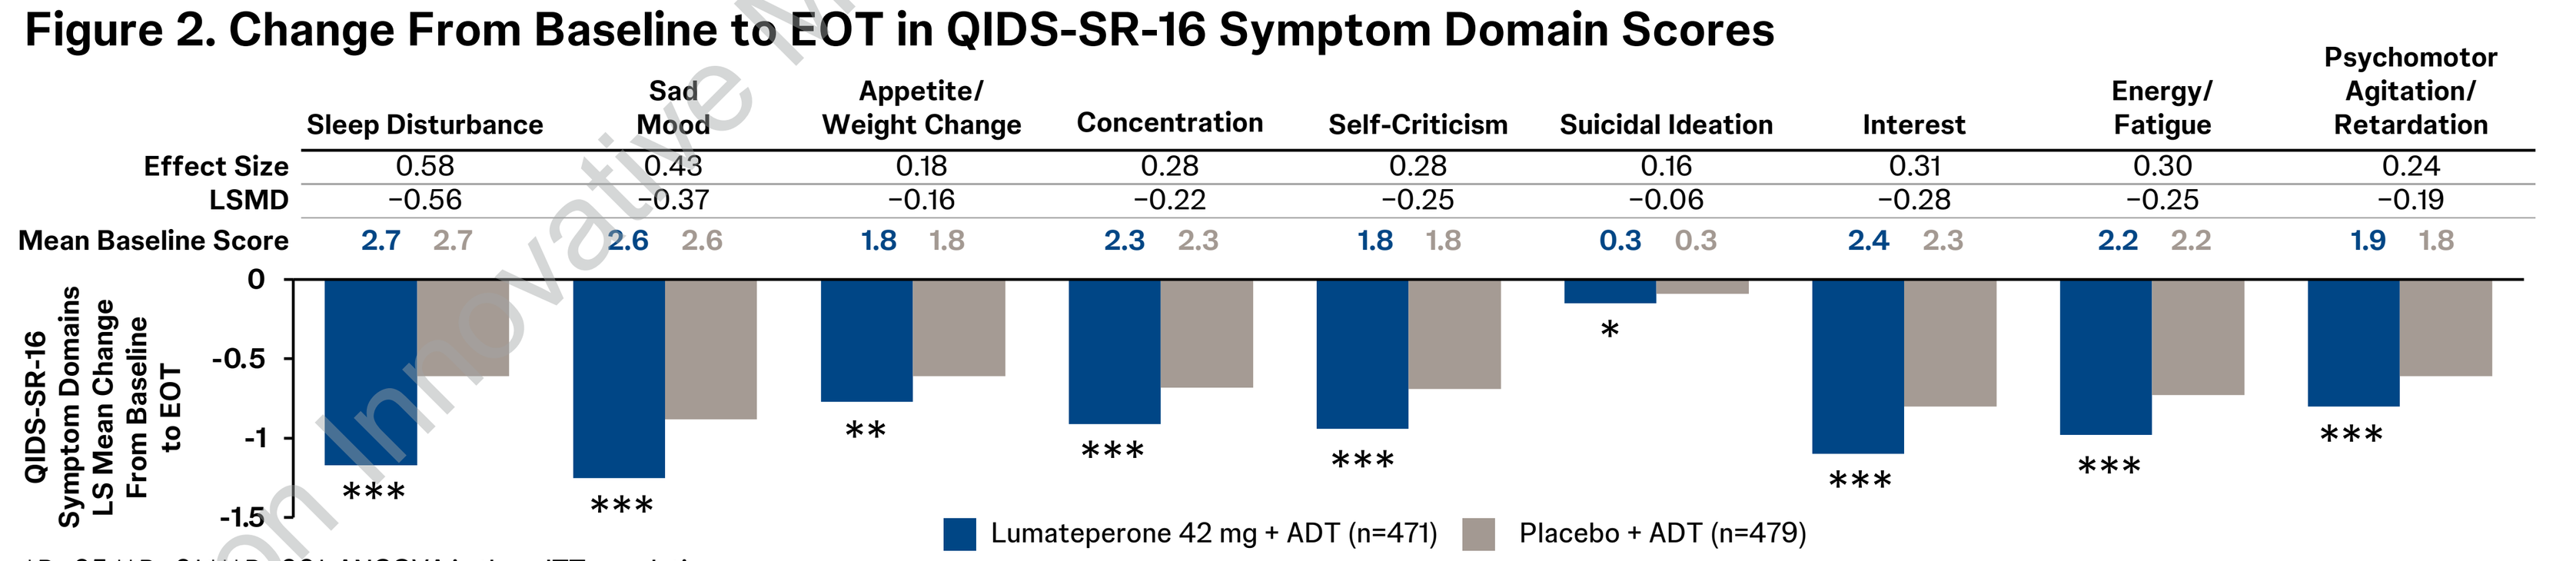
Sleep Disturbance | Sad Mood | Appetite/Weight Change | Concentration | Self-Criticism | Suicidal Ideation | Interest | Energy/Fatigue | Psychomotor Agitation/Retardation
Effect Size 0.58 0.43 0.18 0.28 0.28 0.16 0.31 0.30 0.24
LSMD −0.56 −0.37 −0.16 −0.22 −0.25 −0.06 −0.28 −0.25 −0.19
Mean Baseline Score 2.7 2.7 2.6 2.6 1.8 1.8 2.3 2.3 1.8 1.8 0.3 0.3 2.4 2.3 2.2 2.2 1.9 1.8
QIDS-SR-16 Symptom Domains LS Mean Change From Baseline to EOT
0
-0.5
-1
-1.5
*** *** ** *** *** * *** *** ***
Lumateperone 42 mg + ADT (n=471)
Placebo + ADT (n=479)

*P<.05 **P<.01 ***P<.001. ANCOVA in the mITT population.
ADT, antidepressant therapy; ANCOVA, analysis of covariance; EOT, end of treatment; mITT, modified intent-to-treat; LSMD, least squares mean difference; QIDS-SR-16, Quick Inventory of Depressive Symptomatology-Self Report-16 item.

QIDS-SR-16 Symptom Domains: ≥1- and ≥2-Point Improvement

- A significantly greater percentage of patients receiving lumateperone + ADT versus placebo + ADT achieved ≥1-point improvement on 7 of 9 QIDS-SR-16 symptom domains from baseline to EOT (**Figure 3**)
 - The largest treatment effects were observed for sleep disturbances (odds ratio [OR], 2.76; 95% CI, 2.12-3.61; *P*<.001) and sad mood (OR, 2.32; 95% CI, 1.74-3.10; *P*<.001) (**Figure 3**)

Figure 3. QIDS-SR-16 Symptom Domains: ≥1-Point Improvement From Baseline to EOT

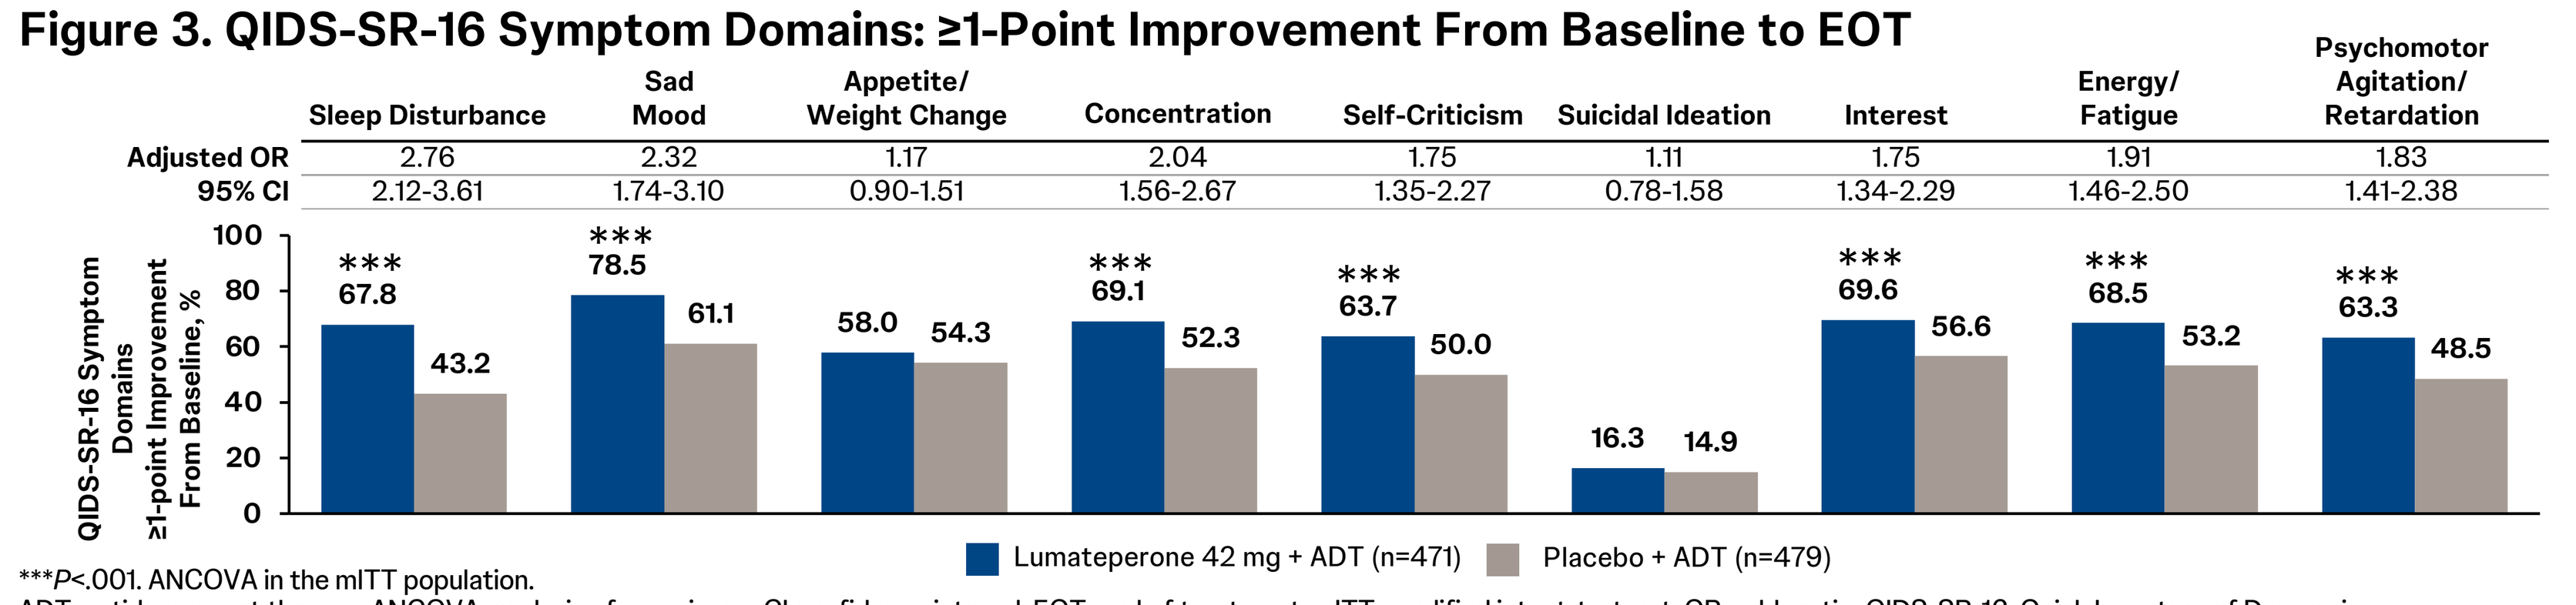
Sleep Disturbance | Sad Mood | Appetite/Weight Change | Concentration | Self-Criticism | Suicidal Ideation | Interest | Energy/Fatigue | Psychomotor Agitation/Retardation
Adjusted OR 2.76 2.32 1.17 2.04 1.75 1.11 1.75 1.91 1.83
95% CI 2.12-3.61 1.74-3.10 0.90-1.51 1.56-2.67 1.35-2.27 0.78-1.58 1.34-2.29 1.46-2.50 1.41-2.38
QIDS-SR-16 Symptom Domains ≥1-point Improvement From Baseline, %
0 20 40 60 80 100
*** 67.8 43.2
*** 78.5 61.1
58.0 54.3
*** 69.1 52.3
*** 63.7 50.0
16.3 14.9
*** 69.6 56.6
*** 68.5 53.2
*** 63.3 48.5
Lumateperone 42 mg + ADT (n=471)
Placebo + ADT (n=479)

***P<.001. ANCOVA in the mITT population.
ADT, antidepressant therapy; ANCOVA, analysis of covariance; CI confidence interval; EOT, end of treatment; mITT, modified intent-to-treat; OR, odds ratio; QIDS-SR-16, Quick Inventory of Depressive Symptomatology-Self Report-16 item.

- A greater proportion of patients treated with lumateperone + ADT achieved ≥2-point improvement across 8 of 9 QIDS-SR-16 symptom domains versus placebo + ADT, with the largest treatment effects observed for sleep disturbance (OR, 2.36; 95% CI, 1.74-3.19; *P*<.001) and sad mood (OR, 2.12; 95% CI, 1.60-2.81; *P*<.001) (**Figure 4**)

Figure 4. QIDS-SR-16 Symptom Domains: ≥2-Point Improvement From Baseline to EOT

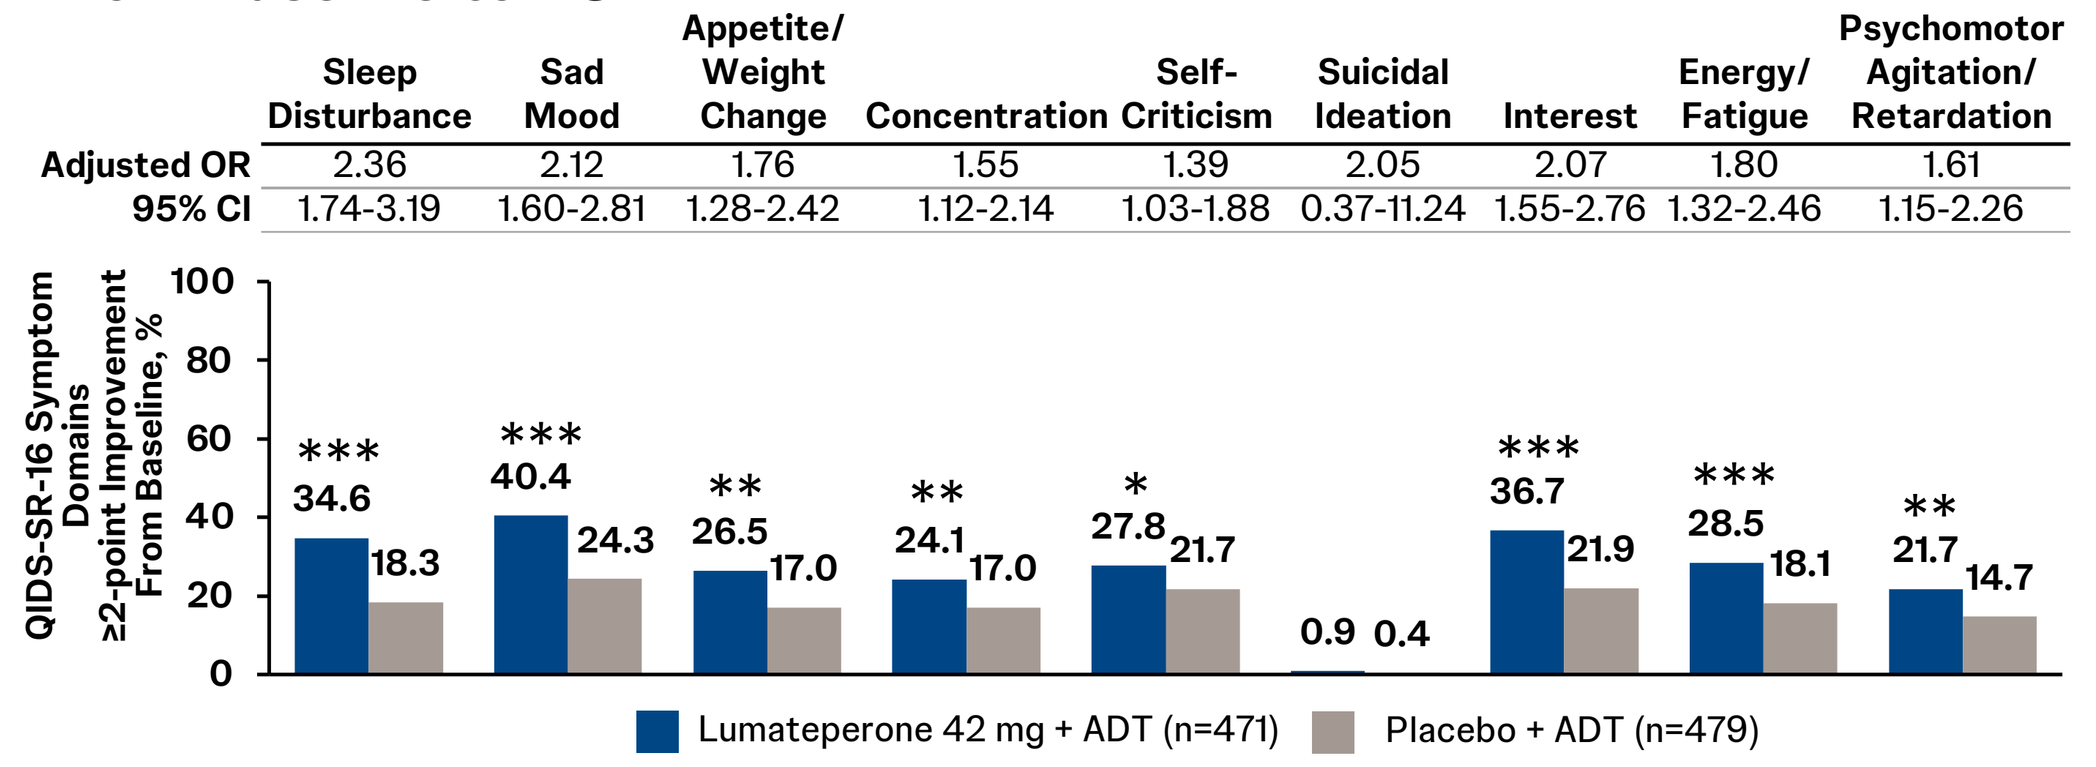
Sleep Disturbance | Sad Mood | Appetite/Weight Change | Concentration | Self-Criticism | Suicidal Ideation | Interest | Energy/Fatigue | Psychomotor Agitation/Retardation
Adjusted OR 2.36 2.12 1.76 1.55 1.39 2.05 2.07 1.80 1.61
95% CI 1.74-3.19 1.60-2.81 1.28-2.42 1.12-2.14 1.03-1.88 0.37-11.24 1.55-2.76 1.32-2.46 1.15-2.26
QIDS-SR-16 Symptom Domains ≥2-point Improvement From Baseline, %
0 20 40 60 80 100
*** 34.6 18.3
*** 40.4 24.3
** 26.5 17.0
** 24.1 17.0
* 27.8 21.7
0.9 0.4
*** 36.7 21.9
*** 28.5 18.1
** 21.7 14.7
Lumateperone 42 mg + ADT (n=471)
Placebo + ADT (n=479)

*P<.05 **P<.01 ***P<.001. ANCOVA in the mITT population.
ADT, antidepressant therapy; ANCOVA, analysis of covariance; CI confidence interval; EOT, end of treatment; mITT, modified intent-to-treat; OR, odds ratio; QIDS-SR-16, Quick Inventory of Depressive Symptomatology-Self Report-16 item.

QIDS-SR-16 Interest-Activity Factor

- Significant improvements were observed in interest-activity factor (LSMD, −0.77; effect size, 0.35; *P*<.001) (**Figure 5**)

Figure 5. Change From Baseline to EOT in QIDS-SR-16 Interest-Activity Factor[a]

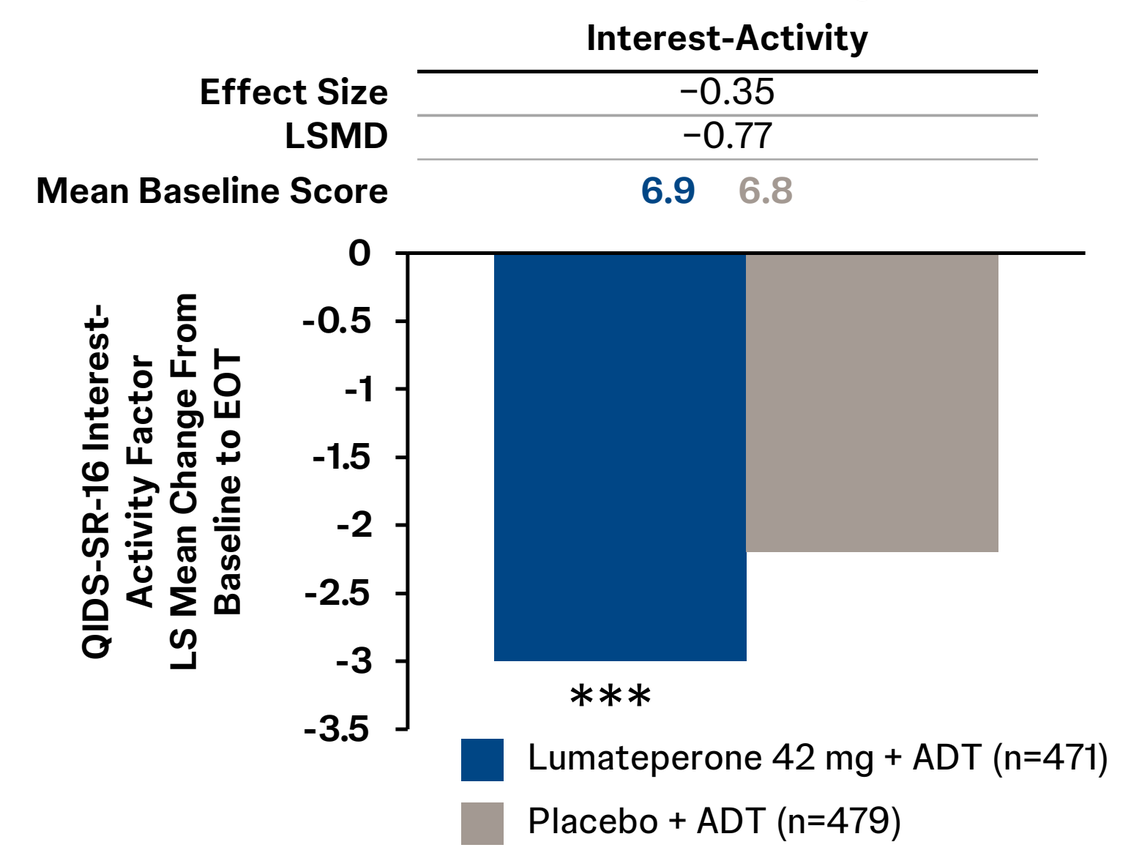
Interest-Activity
Effect Size −0.35
LSMD −0.77
Mean Baseline Score 6.9 6.8
QIDS-SR-16 Interest-Activity Factor LS Mean Change From Baseline to EOT
0 -0.5 -1 -1.5 -2 -2.5 -3 -3.5

Lumateperone 42 mg + ADT (n=471)
Placebo + ADT (n=479)

***P<.001. ANCOVA in the mITT population.
[a]Defined as the sum of QIDS-SR-16 items 10 (concentration/decision-making), 13 (general interest), and 14 (energy level).
ADT, antidepressant treatment; ANCOVA, analysis of covariance; EOT, end of therapy; LSMD, least squares mean difference; mITT, modified intent-to-treat; QIDS-SR-16, Quick Inventory of Depressive Symptomatology-Self Report-16 item.

Conclusions

- In this pooled analysis of 2 short-term studies, adjunctive lumateperone 42 mg significantly improved patient-reported depressive symptoms, as measured by the QIDS-SR-16 Total score, versus placebo + ADT in adults with MDD with inadequate response to ADT
 - These results are in line with the previously reported outcomes with the healthcare professional–assessed MADRS Total score[8,9]
- A greater proportion of patients achieved QIDS-SR-16 response and remission with lumateperone + ADT versus placebo + ADT
- Lumateperone + ADT significantly improved all 9 QIDS-SR-16 symptom domains, with the largest effects observed for sleep disturbance, sad mood, and interest versus placebo + ADT
 - Improvements across the QIDS-SR-16 Total score and symptom domains suggest that lumateperone + ADT may provide broad improvements across multiple dimensions of patient-reported depressive symptoms, with particularly notable effects on sleep disturbance, sad mood, and interest
- A greater proportion of patients treated with lumateperone + ADT achieved both ≥1- and ≥2-point improvements across individual QIDS-SR-16 symptom domains compared with placebo + ADT
- Lumateperone 42 mg + ADT demonstrated improvements in all 3 domains (concentration, interest, and energy/fatigue) of the anhedonia-related QIDS-SR-16 interest-activity factor score

Acknowledgments

The authors thank all study investigators, research staff, and patients for their participation. Medical writing support was provided by Keri Ramessar, PhD, of Nucleus Global, an Inizio company, funded by Intra-Cellular Therapies, a Johnson & Johnson company.

Disclosures

This study was sponsored by Johnson & Johnson.

ME Thase has served as an advisor or a consultant for Autobahn Therapeutics, Axsome Therapeutics, Inc., Clexio Biosciences, Gerson Lehman, GH Therapeutics, H. Lundbeck, A/S, Janssen Pharmaceuticals, Inc., Johnson & Johnson, Luye Pharma Group, Ltd., Merck & Company, Inc., Object Pharma, Otsuka Pharmaceutical Company, Ltd., Pfizer, Inc., Sage Pharmaceuticals, Seelos Pharmaceuticals, and Takeda Pharmaceutical Company, Ltd.; has received grants from Acadia Inc., Alkermes, Axsome Therapeutics Inc., Intra-Cellular Therapies, Inc., Janssen Pharmaceuticals, Inc., Myriad, National Institute of Mental Health, Otsuka Pharmaceutical Company, Ltd., Patient-Centered Outcomes Research Institute (PCORI), and Takeda Pharmaceutical Company, Ltd.; and has received royalties from the American Psychiatric Foundation, Guilford Publications, Herald House, Wolters Kluwer, W.W. Norton & Company, Inc., and spouse's employment with Open Health, which does business with most major pharmaceutical companies. I Turkoz, K Dorsey, and H Zhang are employees of Johnson & Johnson. B Curcio is a contractor of Johnson & Johnson Innovative Medicine R&D. MH Jha has received contract research grants from Neurocrine Bioscience, Navitor/Supernus and Janssen Research & Development; honorarium to serve as Section Editor of the Psychiatry & Behavioral Health Learning Network and as Guest Editor for Psychiatric Clinics of North America from Elsevier; consultant fees from Janssen Scientific Affairs, Sanofi, Neurocrine, AbbVie, MindMed, and Boehringer Ingelheim; fees to serve on a data safety and monitoring board for Worldwide Clinical Trials (Eliem, Skye and Inversargo), Vicore Pharma, and IQVIA (Click); and honoraria for educational presentations from North American Center for Continuing Medical Education, Medscape/WebMD, Clinical Care Options, Soterix Medical Inc., Physicians' Education Resource, Efficient CME, and H.C. Wainwright & Co.

Neuropsychiatry

Scan the QR code
The QR code is intended to provide scientific information for individual reference, and the information should not be altered or reproduced in any way

Email questions to Manish K. Jha at Manish.Jha@UTSouthwestern.edu

Presented at Psych Congress 2026, September 15-19, 2026, New Orleans, LA, USA

References:

1. Su YA, Si T. *Gen Psychiatry*. 2022;35:e100724; 2. Malhi GS, Mann JJ. *Lancet*. 2018;392:2299-2312; 3. Rush AJ, et al. *Biol Psychiatry*. 2006;59:493-501; 4. Palmer EOC, et al. *Neuropsychiatr Dis Treat*. 2024;20:671-687; 5. Rush AJ, et al. *Biol Psychiatry*. 2003;54:573-583; 6. Caplyta (lumateperone). Prescribing information. Intra-Cellular Therapies, Inc; 2026; 7. Davis RE, Correll CU. *Expert Rev Neurother*. 2016;16:601-614; 8. Durgam S, et al. *J Clin Psychiatry*. 2025;86:25m15848; 9. Durgam S, et al. *Am J Psychiatry*. 2025;182:1072-1082; 10. Durgam S, et al. Presented at the 36th World Congress of Neuropsychopharmacology/9th ASCNP Congress (CINP-AsCNP) Joint Congress, June 15-18, 2025; Melbourne, Australia. Oral and poster presentation.