Lumateperone for the Prevention of Relapse in Patients with Schizophrenia: Results From a Double-Blind, Placebo-Controlled, Randomized Withdrawal, Phase 3 Trial

Suresh Durgam, MD[1]; Willie R. Earley, MD[1]; Susan G. Kozauer, MD[1*]; Jason Huo, PhD[1]; Hassan Lakkis, PhD[1]; Christopher Gallardo, PhD[1]; Christoph U. Correll, MD[2-5]

[1] Intra-Cellular Therapies, a Johnson & Johnson Company, Bedminster, NJ, USA; [2] Department of Psychiatry, The Zucker Hillside Hospital, Northwell Health, Glen Oaks, NY, USA; [3] Department of Psychiatry and Molecular Medicine, Zucker School of Medicine at Hofstra/Northwell, Hempstead, NY, USA; [4] Department of Child and Adolescent Psychiatry, Charité Universitätsmedizin Berlin, Berlin, Germany; [5] German Center for Mental Health (DZPG), partner site Berlin, Berlin, Germany; *Former employee

This material is distributed for scientific purposes on J&J Medical Connect, and is not for promotional use

BACKGROUND

- Relapse is common in patients with schizophrenia and is associated with worsening symptoms, cognitive deterioration, poorer quality of life, and functional impairment[1]
- Psychosocial outcomes and quality of life further decline with increasing frequency of relapse[2]
- Although maintenance antipsychotic treatment has shown efficacy in reducing relapse rate in patients with schizophrenia, current treatment options can be limited by adverse effects such as movement disorders and weight gain that may lead to nonadherence[3]
 - Thus, a novel antipsychotic treatment that helps prevent relapse with a tolerable safety profile may lead to long-term benefits for patients with schizophrenia[2]
- Lumateperone is a mechanistically novel US Food and Drug Administration–approved antipsychotic to treat schizophrenia and depressive episodes associated with bipolar I or bipolar II disorder as monotherapy and as adjunctive therapy with lithium or valproate[4,5]
 - Lumateperone is a simultaneous modulator of serotonin, dopamine, and glutamate neurotransmission[5]
 - Specifically, lumateperone is a potent serotonin 5-HT_{2A} receptor antagonist, a dopamine D_2 receptor presynaptic partial agonist and postsynaptic antagonist, a D_1 receptor–dependent indirect modulator of glutamatergic AMPA and NMDA currents, and a serotonin reuptake inhibitor[5]
 - This novel mechanism of action with multimodal effects may confer robust efficacy with improved tolerability compared with current treatment options
- This Phase 3, multicenter, multinational, double-blind, placebo-controlled, randomized withdrawal trial (Study 304, NCT04959032) investigated the efficacy and safety of lumateperone 42 mg for the prevention of relapse in adult patients with schizophrenia

METHODS

- Adults with schizophrenia experiencing a current psychotic episode received open-label, oral lumateperone 42 mg treatment once daily for 18 weeks
 - Patients who achieved stability by the end of the 6-week run-in period and were still stable at Week 18 were randomized 1:1 to double-blind treatment with lumateperone 42 mg or placebo for 26 weeks or until relapse (**Figure 1**)

Figure 1. Study Design

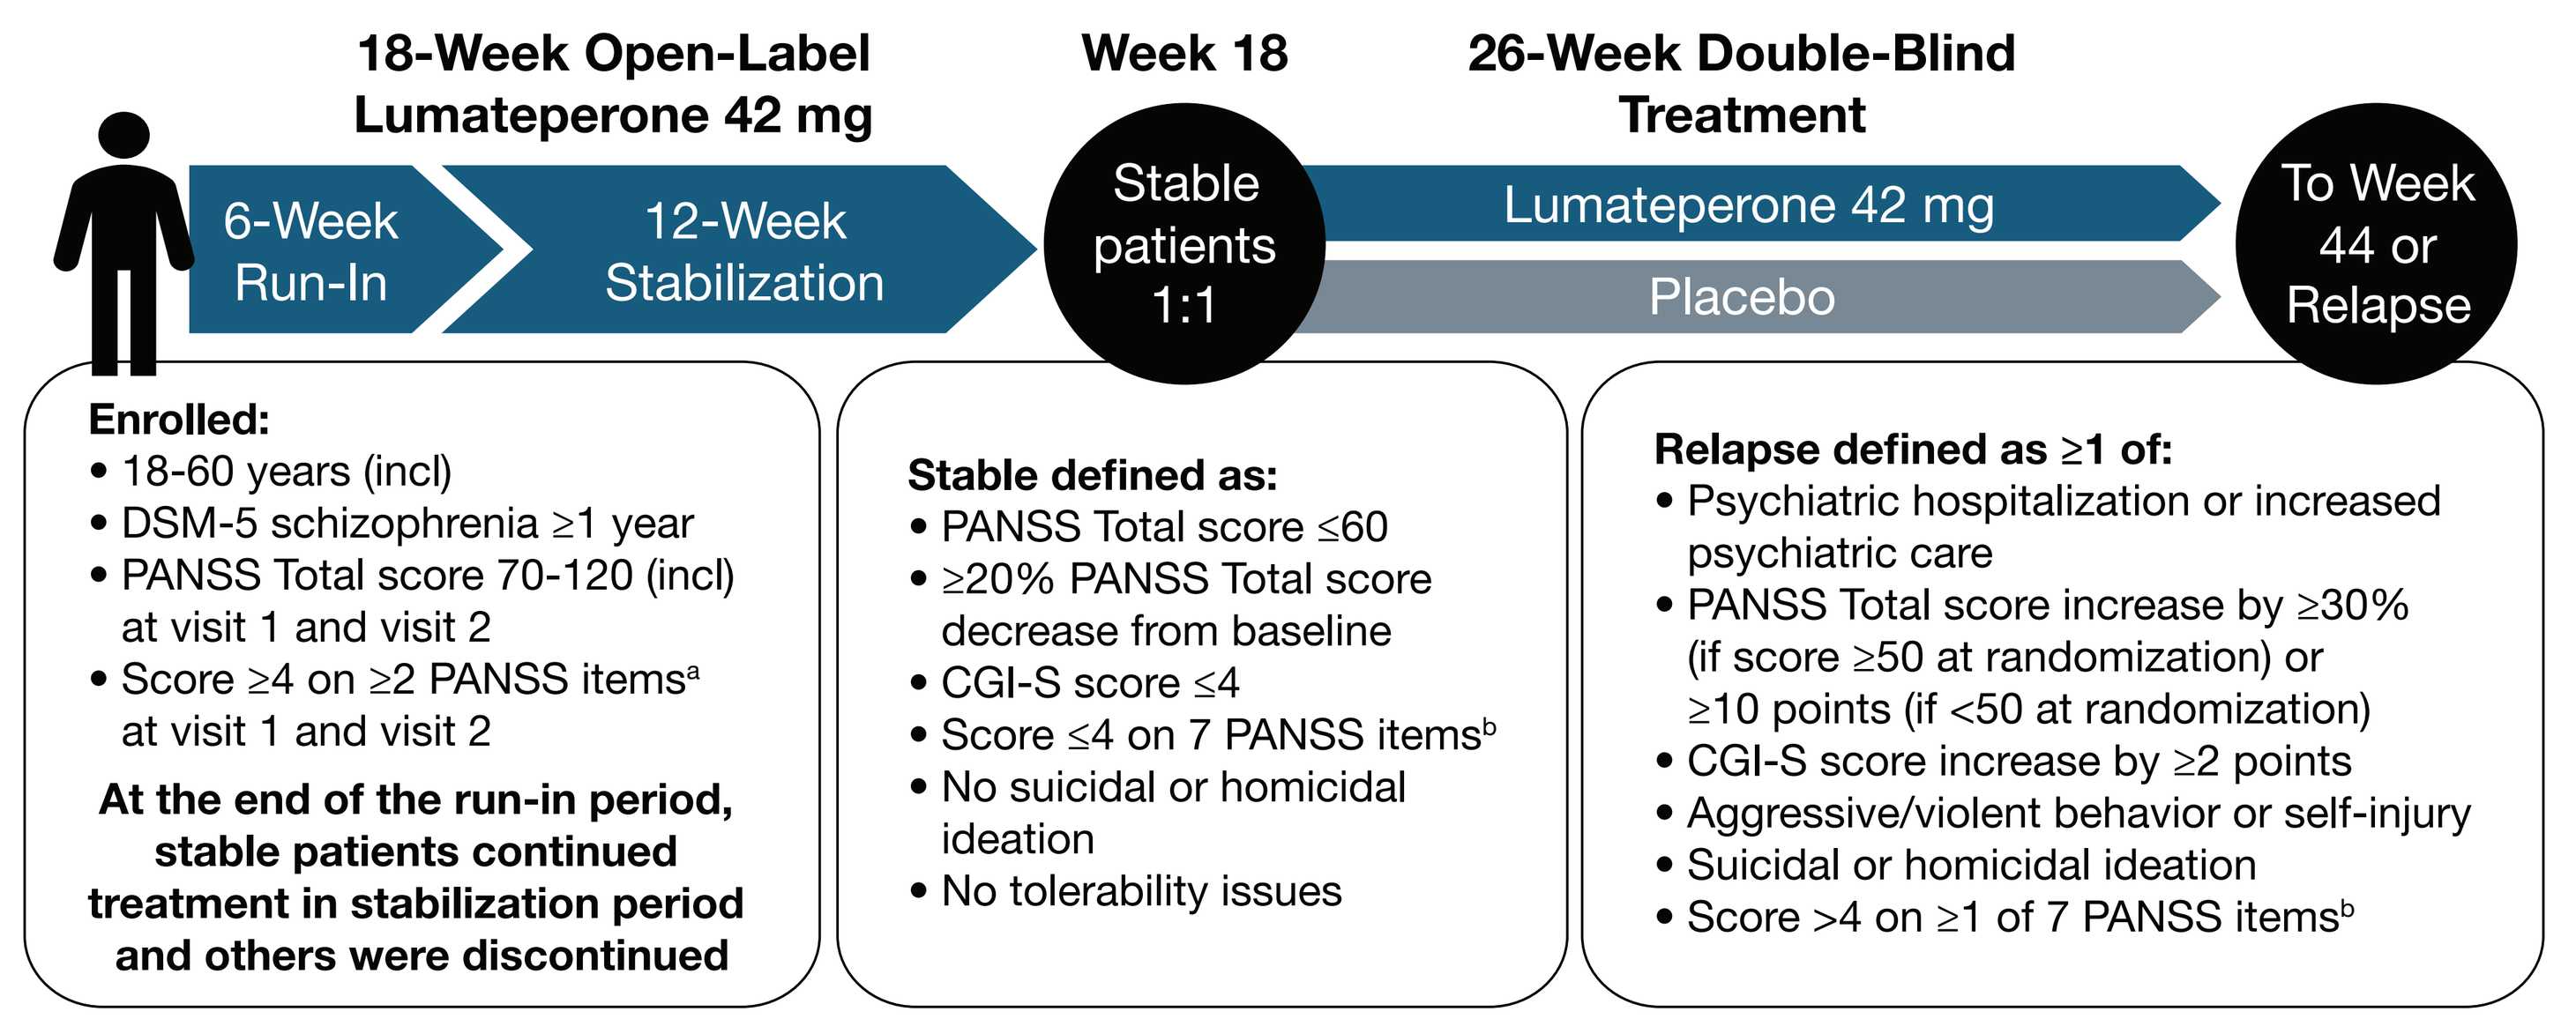
18-Week Open-Label Lumateperone 42 mg
Week 18
26-Week Double-Blind Treatment
6-Week Run-In
12-Week Stabilization
Stable patients 1:1
Lumateperone 42 mg
Placebo
To Week 44 or Relapse
Enrolled:
• 18-60 years (incl)
• DSM-5 schizophrenia ≥1 year
• PANSS Total score 70-120 (incl) at visit 1 and visit 2
• Score ≥4 on ≥2 PANSS items[a] at visit 1 and visit 2
At the end of the run-in period, stable patients continued treatment in stabilization period and others were discontinued
Stable defined as:
• PANSS Total score ≤60
• ≥20% PANSS Total score decrease from baseline
• CGI-S score ≤4
• Score ≤4 on 7 PANSS items[b]
• No suicidal or homicidal ideation
• No tolerability issues
Relapse defined as ≥1 of:
• Psychiatric hospitalization or increased psychiatric care
• PANSS Total score increase by ≥30% (if score ≥50 at randomization) or ≥10 points (if <50 at randomization)
• CGI-S score increase by ≥2 points
• Aggressive/violent behavior or self-injury
• Suicidal or homicidal ideation
• Score >4 on ≥1 of 7 PANSS items[b]

[a] PANSS items P1 (delusions), P3 (hallucinatory behavior), P2 (conceptual disorganization), and P6 (suspiciousness/persecution). [b] PANSS items P1 (delusions), P2 (conceptual disorganization), P3 (hallucinatory behavior), P6 (suspiciousness), P7 (hostility), G8 (uncooperativeness) and G14 (poor impulse control).
CGI-S, Clinical Global Impression Scale-Severity; DSM-5, *Diagnostic and Statistical Manual of Mental Disorders*, 5th edition; incl, inclusive; PANSS, Positive and Negative Syndrome Scale.

- The primary endpoint was the time to first symptom relapse during double-blind treatment
- The key secondary endpoint was time to all-cause discontinuation (including relapse) during double-blind treatment
- Safety assessments included adverse events (AEs), body morphology, laboratory parameters, extrapyramidal symptoms (EPS), and suicidality

RESULTS

Patient Population

- Of 866 patients enrolled, 592 were in the open-label safety population and 228 (39%) patients completed the open-label lumateperone treatment period
 - A total of 228 patients were stable at Week 18 and randomized in the double-blind treatment phase, 224 were treated and included in the safety and intent-to-treat populations (lumateperone, 110; placebo, 114)
 - Of patients in the double-blind treatment phase, 130 (58%) completed double-blind treatment without relapse
- Demographics and baseline characteristics were generally similar between groups (**Table 1**)
 - Compared with the population in the open-label treatment phase, a greater proportion of White patients continued to the double-blind treatment phase
 - As expected, mean Positive and Negative Syndrome Scale (PANSS) Total scores improved from the open-label treatment period baseline (91.5) to the double-blind treatment period baseline (lumateperone, 50.8; placebo, 51.3)

Table 1. Baseline Demographics and Disease Characteristics (Safety Population)

	Open-Label Treatment	Double-Blind Treatment	
	Lumateperone 42 mg (n=592)	Lumateperone 42 mg (n=110)	Placebo (n=114)
Age, mean (range), years	42.5 (18-60)	44.9 (23-60)	45.4 (25-60)
Sex, n (%)			
Male	414 (69.9)	73 (66.4)	70 (61.4)
Female	178 (30.1)	37 (33.6)	44 (38.6)
Race, n (%)			
White	299 (50.5)	77 (70.0)	84 (73.7)
Black	277 (46.8)	31 (28.2)	29 (25.4)
Asian	8 (1.4)	2 (1.8)	0
Other	8 (1.4)	0	1 (0.9)
Hispanic or Latino ethnicity, n (%)	45 (7.6)	4 (3.6)	6 (5.3)
No. previous psychiatric hospitalizations, mean (range)	5.0 (0-45)	4.9 (0-27)	5.6 (0-26)

Efficacy

- Lumateperone met the primary endpoint, significantly delaying the time to relapse vs placebo during double-blind treatment (**Figure 2**)
- Fewer relapses occurred with lumateperone (18 [16.4%]) than placebo (44 [38.6%]), corresponding to a number needed to treat (NNT) of 5
 - The most common reason for relapse in both groups was PANSS Total score increase (lumateperone, 17 [15.5%]; placebo, 44 [38.6%]), followed by score >4 on ≥1 of 7 PANSS items (lumateperone, 12 [10.9%]; placebo, 19 [16.7%]) and ≥2 point CGI-S score increase in the Visit 14 score (lumateperone, 9 [8.2%]; placebo, 28 [24.6%])

Figure 2. Cumulative Rate of Relapse During Double-Blind Treatment (ITT Population)

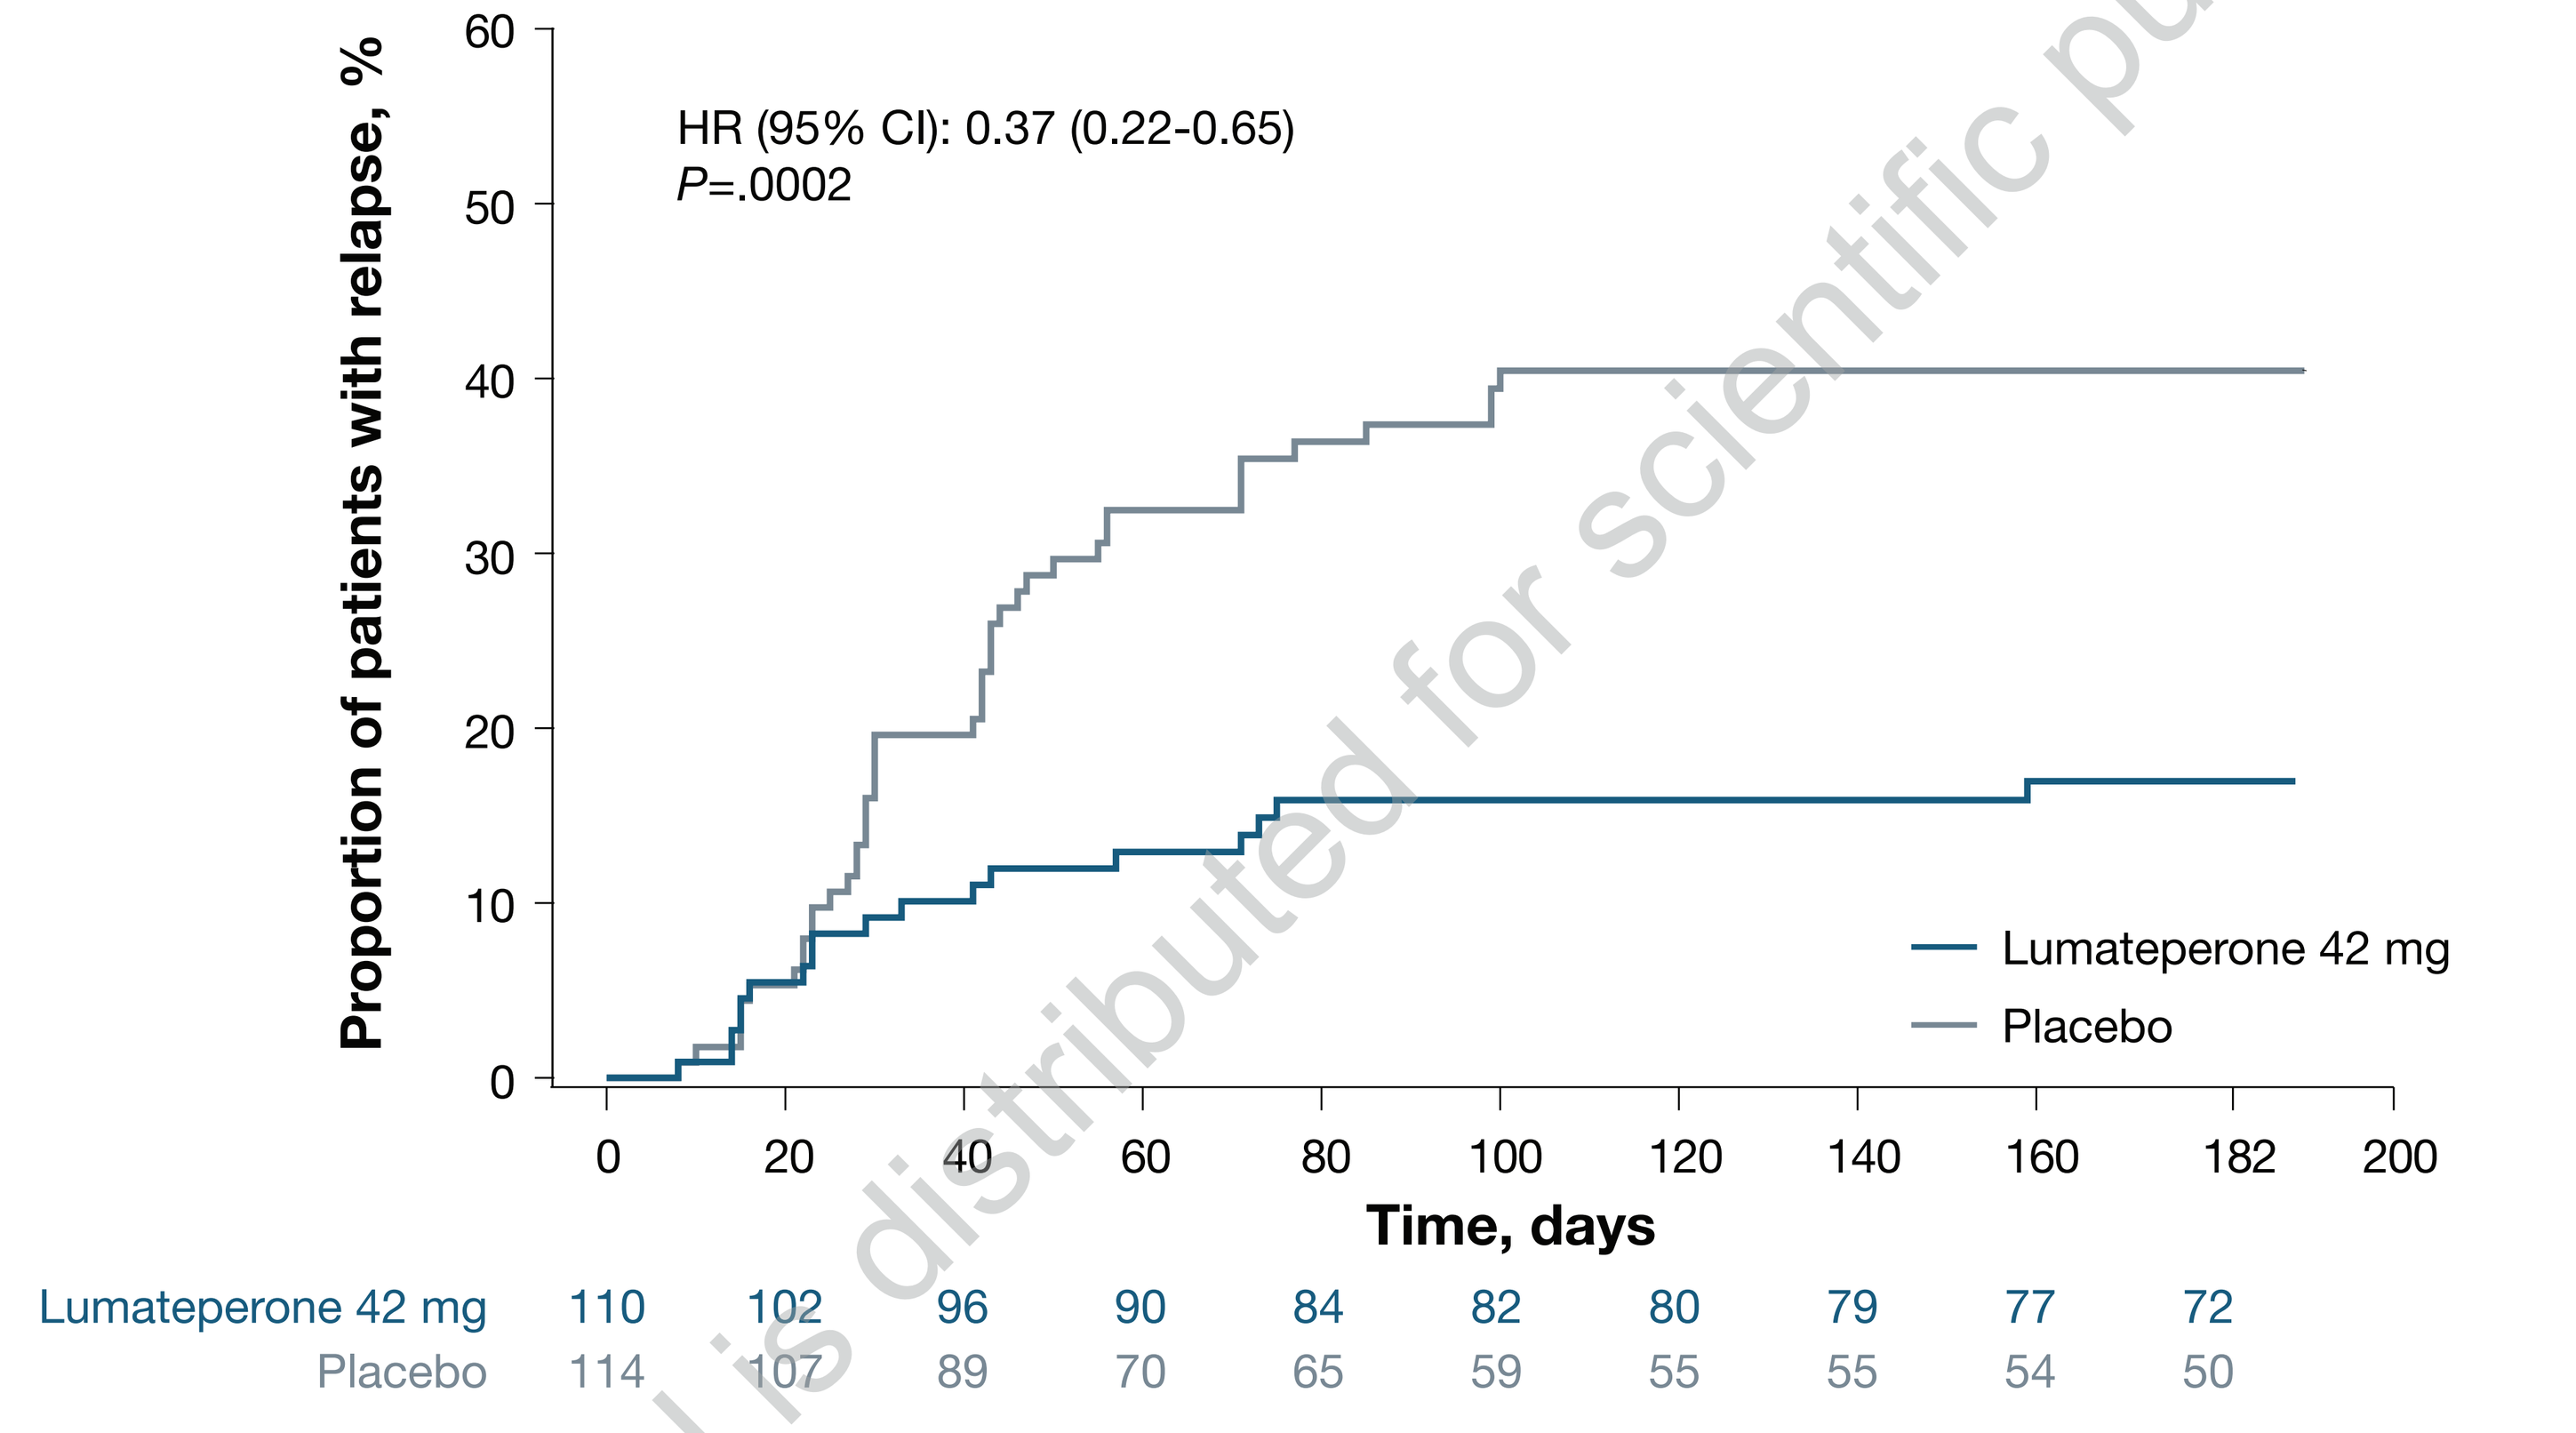
HR (95% CI): 0.37 (0.22-0.65)
P=.0002
Proportion of patients with relapse, %
0
10
20
30
40
50
60
Lumateperone 42 mg
Placebo
0 20 40 60 80 100 120 140 160 182 200
Time, days
Lumateperone 42 mg 110 102 96 90 84 82 80 79 77 72
Placebo 114 107 89 70 65 59 55 55 54 50

HR, hazard ratio; ITT, intent-to-treat.
P value for prespecified primary analysis using the log-rank test. Estimates of HR and 95% CI based on the Cox proportional hazards model with treatment group as the explanatory variable.

- Lumateperone treatment also significantly delayed time to all-cause discontinuation (including relapse) compared with placebo (**Figure 3**)
 - The rate of all-cause discontinuation was lower with lumateperone (34 [30.9%]) than placebo (60 [52.6%]) corresponding to an NNT of 5

Figure 3. Cumulative Rate of All Cause Discontinuation During Double-Blind Treatment (ITT Population)

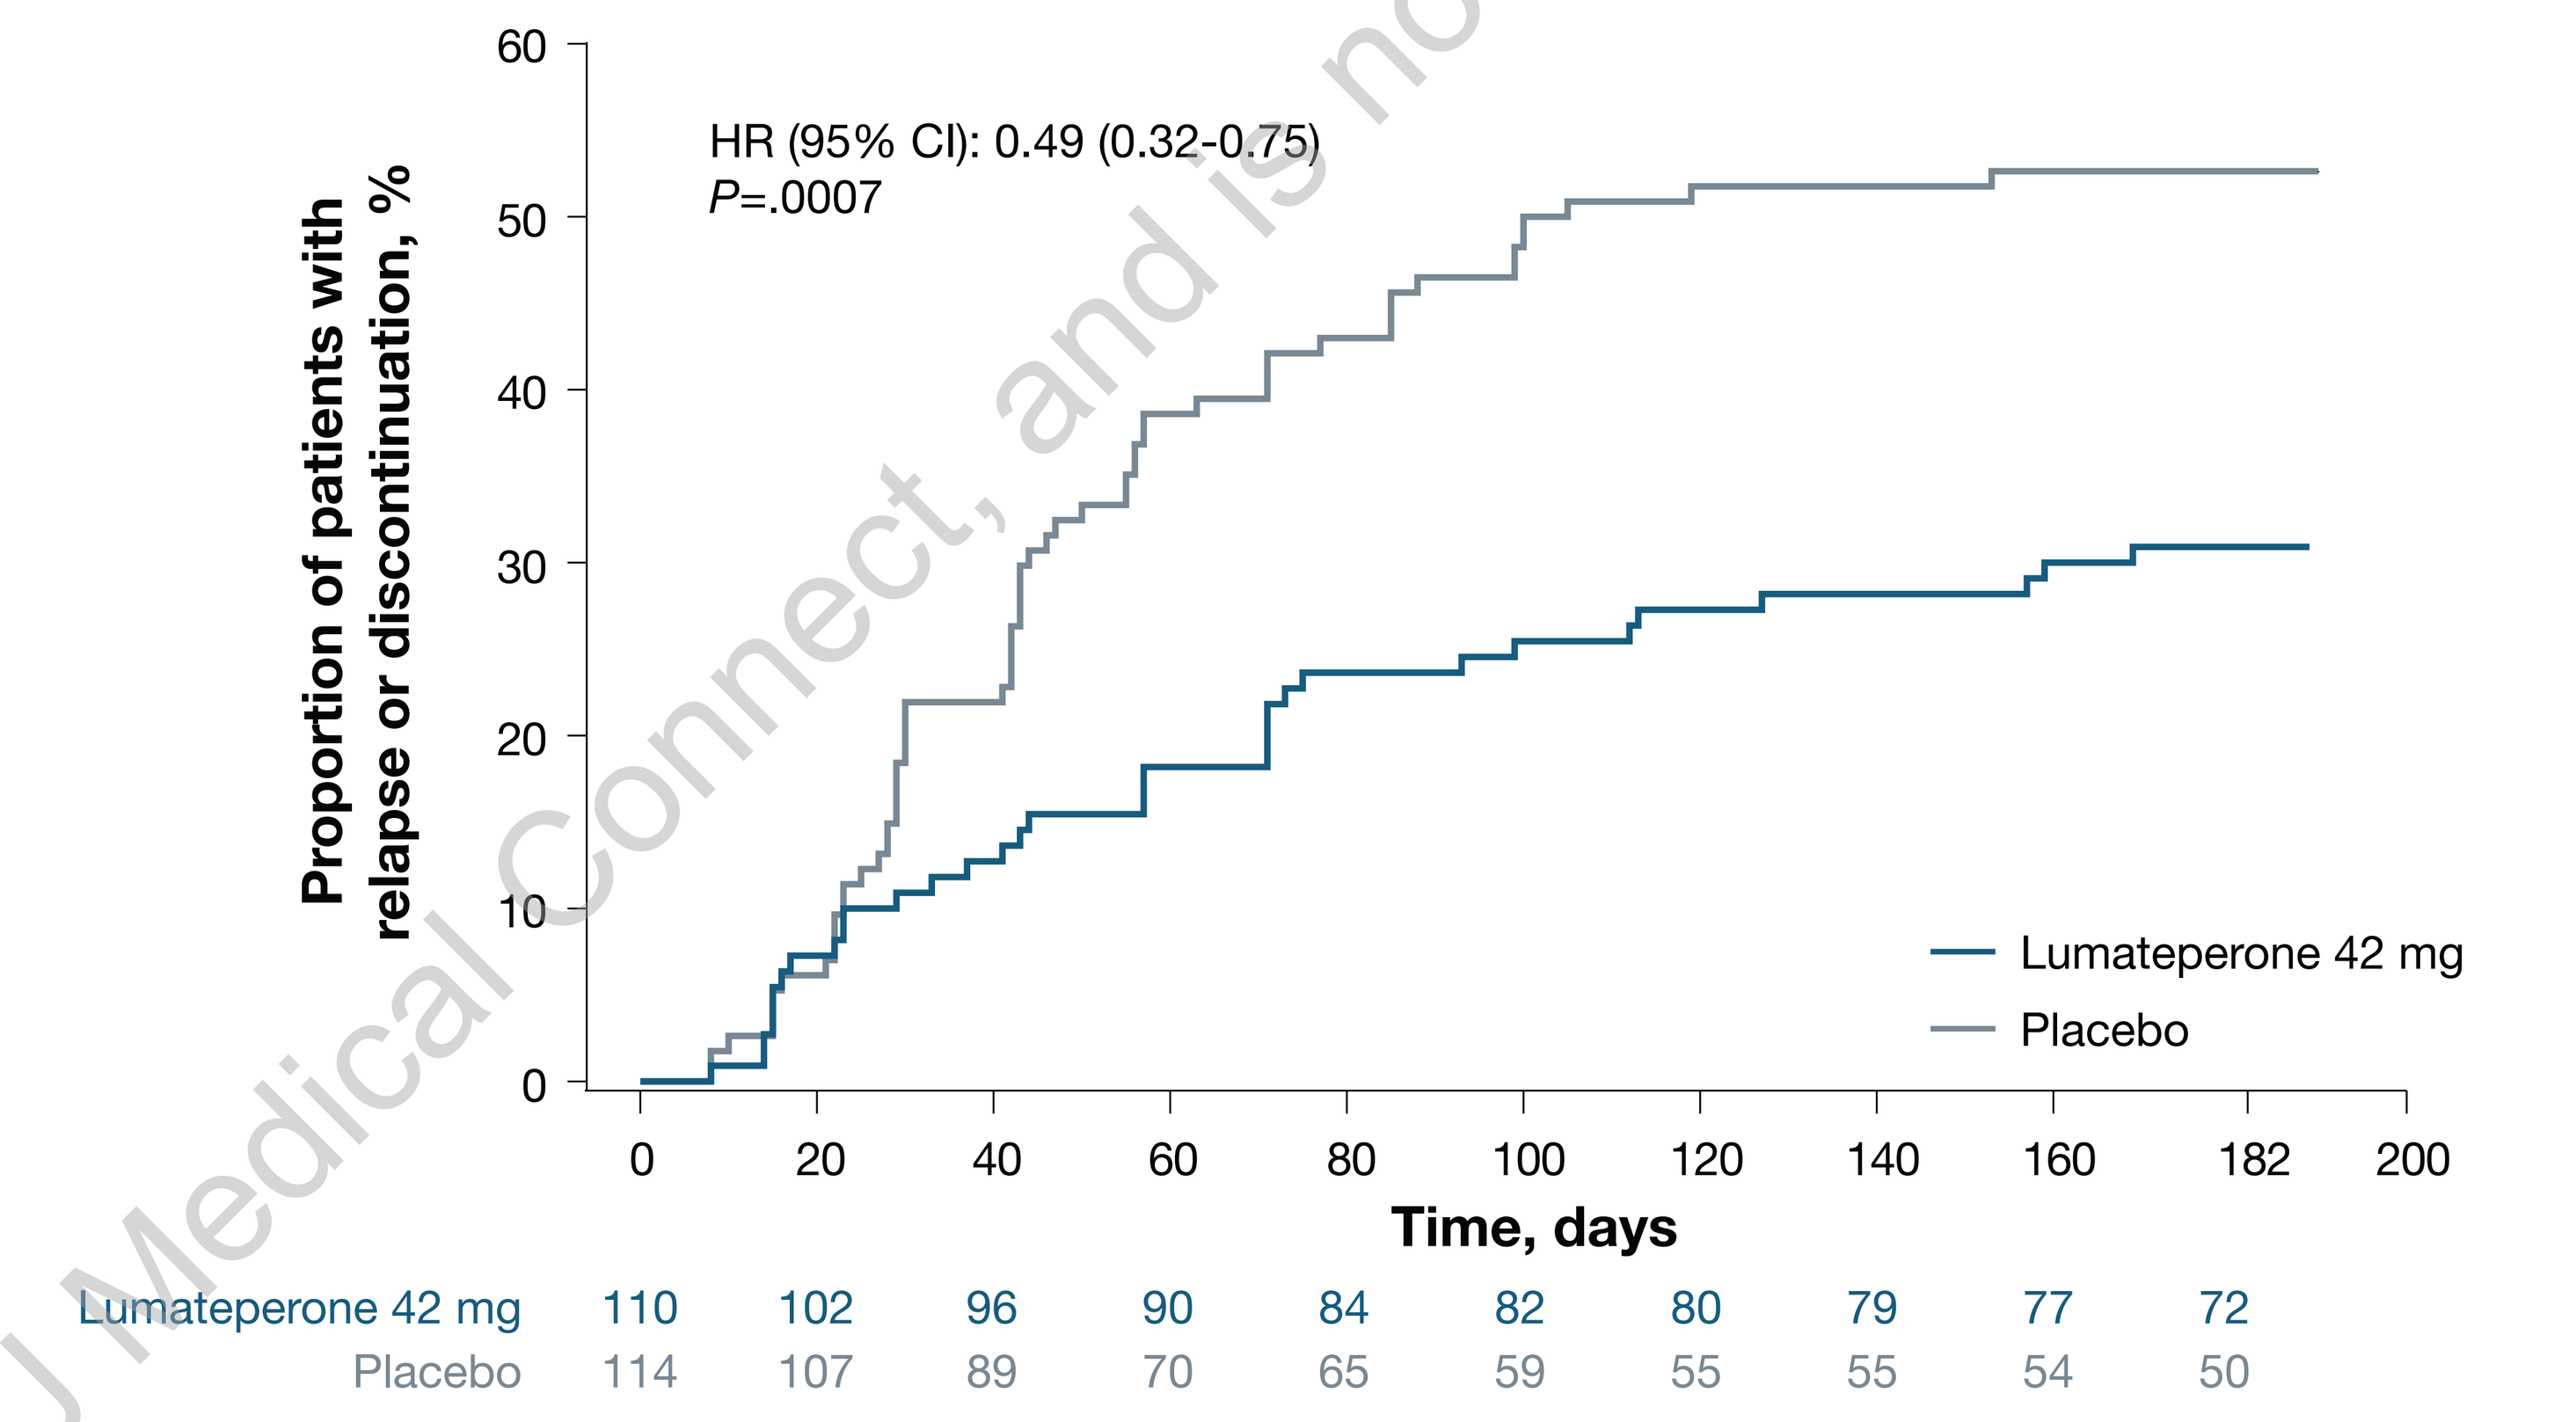
HR (95% CI): 0.49 (0.32-0.75)
P=.0007
Proportion of patients with relapse or discontinuation, %
0
10
20
30
40
50
60
Lumateperone 42 mg
Placebo
0 20 40 60 80 100 120 140 160 182 200
Time, days
Lumateperone 42 mg 110 102 96 90 84 82 80 79 77 72
Placebo 114 107 89 70 65 59 55 55 54 50

HR, hazard ratio; ITT, intent-to-treat.
P value based on log-rank test. Estimates of HR and 95% CI based on the Cox proportional hazards regression model with treatment group as the explanatory variable.

Safety

- During the double-blind treatment period mean treatment duration was 144.5 days for the lumateperone group and 111.9 days for the placebo group
- Treatment-emergent AEs (TEAEs) occurred in 52% of patients in the open-label treatment period and in similar frequencies in the lumateperone (33.6%) and placebo (30.7%) groups in the double-blind treatment period (**Table 2**)
 - The most common TEAE was headache in both the open-label and double-blind treatment periods
 - For most patients, TEAEs were mild or moderate in severity
- The rate of EPS-related TEAEs was low and similar to placebo during double-blind treatment (**Table 2**)
 - There were no notable changes from baseline in EPS during the study as assessed by the Abnormal Involuntary Movement Scale, Barnes Akathisia Rating Scale, and Simpson-Angus Scale

Table 2. Adverse Events Summary (Safety Population)

	Open-Label Treatment	Double-Blind Treatment	
Patients, n (%)	Lumateperone 42 mg (n=592)	Lumateperone 42 mg (n=110)	Placebo (n=114)
≥1 TEAE	308 (52.0)	37 (33.6)	35 (30.7)
Drug-related TEAE	144 (24.3)	10 (9.1)	10 (8.8)
Discontinued treatment due to AE	52 (8.8)	2 (1.8)	2 (1.8)
SAE	23 (3.9)	1 (0.9)	7 (6.1)
Death	1 (0.2)[a]	0	0
TEAEs in ≥5% of the lumateperone group[b]			
Headache	78 (13.2)	9 (8.2)	4 (3.5)
EPS-related TEAEs[c]	16 (2.7)	1 (0.9)	1 (0.9)

[a] The cause of death was suicide, unrelated to study drug. [b] And at more than twice the rate of placebo in the double-blind treatment population. [c] According to the standard Medical Dictionary for Regulatory Activities query narrow criteria for EPS-related TEAEs.
EPS, extrapyramidal symptom; SAE, serious adverse event; TEAE, treatment-emergent adverse event.

- Body morphology parameters remained stable with lumateperone from the baseline of the open-label treatment period to the end of the double-blind treatment period (**Table 3**)
 - Potentially clinically significant weight increase and decrease (≥7% change from open-label treatment baseline) occurred at similar rates in the lumateperone group (11.8% increase and 13.6% decrease) and placebo group (both 9.7%) at the end of double-blind treatment
- There were no clinically relevant increases at the end of the double-blind treatment period in prolactin or cardiometabolic parameters (**Table 3**)
- Based on the Columbia-Suicide Severity Rating Scale, suicidal behavior was rare (open-label treatment, 0.3%; double-blind treatment, 0%)
 - Emergence of suicidal ideation was low during open-label treatment (2.3%) and double-blind treatment (lumateperone, 0.9%; placebo, 0%)

Table 3. Mean Change From Baseline in Body Morphology, Prolactin, and Cardiometabolic Parameters During Double-Blind Treatment (Safety Population)

	Double-Blind Treatment			
	Lumateperone 42 mg (n=110)		Placebo (n=114)	
	Baseline Mean (SD)[a]	Mean Change at EOT (SD)[b]	Baseline Mean (SD)[a]	Mean Change at EOT (SD)[b]
Weight, kg	80.5 (18.08)	0.2 (5.23)	82.9 (19.32)	–0.1 (4.14)
BMI, kg/m²	27.0 (5.35)	0.1 (1.76)	27.8 (5.52)	0.0 (1.47)
Waist circumference, cm	95.2 (13.59)	0.4 (5.16)	95.5 (13.97)	–1.1 (5.34)
Prolactin, ng/mL	17.8 (18.58)	–3.3 (20.01)	15.6 (18.29)	–2.9 (14.44)
Cholesterol, mg/dL				
Total	184.5 (39.15)	–6.1 (37.32)	189.1 (43.03)	–0.8 (40.10)
HDL	47.6 (11.08)	2.0 (12.26)	47.2 (13.04)	4.9 (15.25)
LDL	127.9 (39.90)	–4.8 (35.06)	132.0 (43.03)	–2.9 (41.04)
Triglycerides, mg/dL	149.4 (87.28)	–11.0 (83.88)	154.5 (119.74)	–3.3 (132.86)
Glucose, mg/dL	91.3 (12.68)	2.0 (15.73)	97.5 (42.78)	–2.3 (37.99)
Insulin, mIU/L	15.9 (19.78)	3.2 (31.61)	17.3 (25.46)	2.6 (40.37)

[a] Baseline was defined as the last assessment before the first dose of open-label study drug. bMean change from baseline of open-label treatment to end of double-blind treatment.
BMI, body mass index; EOT, end of treatment; HDL, high-density lipoprotein; LDL, low-density lipoprotein.

CONCLUSIONS

- **Lumateperone 42 mg demonstrated efficacy as a maintenance treatment in patients with schizophrenia, significantly delaying the time to relapse compared with placebo (*P*=.0002)**
 - **The relative risk of time to relapse with lumateperone 42 mg was reduced by 63% compared with placebo (HR 0.37)**
- **Lumateperone 42 mg also reduced absolute relapse rates compared with placebo, translating to an NNT of 5**
- **Lumateperone 42 mg was generally safe and well tolerated with minimal EPS risk and no notable changes in prolactin or cardiometabolic parameters, consistent with prior lumateperone trials**
- **These results support the benefit of continued long-term treatment with lumateperone 42 mg in adults with schizophrenia**

REFERENCES

1. Olivares JM, et al. *Ann Gen Psychiatry.* 2013;12(1):32.
2. Lin D, et al. *Front Psychiatry.* 2021;12:695672.
3. Ceraso A, et al. *Cochrane Database Syst Rev.* 2020;8(8):CD008016.
4. Caplyta. Prescribing information. Intra-Cellular Therapies, Inc.;2023.
5. Titulaer J, et al. *Eur Neuropsychopharmacol.* 2022;62:22-35.

DISCLOSURES AND ACKNOWLEDGMENTS

S Durgam, WR Earley, J Huo, H Lakkis, and C Gallardo are full-time employees of Intra-Cellular Therapies, a Johnson & Johnson Company. SG Kozauer is a former employee of Intra-Cellular Therapies, a Johnson & Johnson Company.

CU Correll has been a consultant and/or advisor to or has received honoraria from AbbVie, Acadia, Adock Ingram, Alkermes, Allergan, Angelini, Aristo, Biogen, Boehringer-Ingelheim, Bristol-Meyers Squibb, Cardio Diagnostics, Cerevel, CNX Therapeutics, Compass Pathways, Darnitsa, Delpor, Denovo, Eli Lilly, Gedeon Richter, Hikma, Holmusk, IntraCellular Therapies Inc., Jamjoom Pharma, Janssen/J&J, Karuna, LB Pharma, Lundbeck, MedInCell, MedLink, Merck, Mindpax, Mitsubishi Tanabe Pharma, Maplight, Mylan, Neumora Therapeutics, Neurocrine, Neurelis, Newron, Noven, Novo Nordisk, Otsuka, PPD Biotech, Recordati, Relmada, Reviva, Rovi, Sage, Saladax, Sanofi, Seqirus, SK Life Science, Sumitomo Pharma America, Sunovion, Sun Pharma, Supernus, Tabuk, Takeda, Teva, Terran, Tolmar, Vertex, Viatris and Xenon Pharmaceuticals. He provided expert testimony for Janssen, Lundbeck, and Otsuka. He served on a Data Safety Monitoring Board for Compass Pathways, Denovo, IntraCellular Therapies Inc., Lundbeck, Relmada, Reviva, Rovi, Supernus, and Teva. He has received grant support from Boehringer-Ingelheim, Janssen, and Takeda. He received royalties from UpToDate and is also a stock option holder of Cardio Diagnostics, Kuleon Biosciences, LB Pharma, Medlink, Mindpax, Quantic, and Terran.

Key contributors: All authors: data analysis/interpretation and writing/critical review. Additionally, S Durgam, WR Earley, SG Kozauer, J Huo, and H Lakkis participated in study design/conduct.

The authors thank all study investigators, research staff, and patients for their participation. Medical writing support was provided by Kendall Foote, PhD, of Nucleus Global, an Inizio company, funded by Intra-Cellular Therapies, a Johnson & Johnson Company.

Scan the QR code for the full digital poster. The QR code is intended to provide scientific information for individual reference, and the information should not be altered or reproduced in any way.

Presented at the Neuroscience Education Institute Fall Congress, November 6-9, 2025, Colorado Springs, CO

Email questions to Christopher Gallardo at cgallardo@itci-inc.com