Long-Term Adjunctive Lumateperone Treatment in Major Depressive Disorder: Results From a Six-Month Open-Label Extension Study

Willie R. Earley, MD[1]; Suresh Durgam, MD[1]; Susan G. Kozauer, MD[1*]; Changzheng Chen, PhD[1]; Francis Brennan, PhD[1]; Andrew J. Cutler, MD[2]

[1] Intra-Cellular Therapies, a Johnson & Johnson Company, Bedminster, NJ, USA; [2] Department of Psychiatry, SUNY Upstate Medical University, Lakewood Ranch, FL, USA; *Former employee

Intra-Cellular THERAPIES

This material is distributed for scientific purposes on J&J Medical Connect, and is not for promotional use

BACKGROUND

- Major depressive disorder (MDD) is a highly burdensome illness that is associated with functional impairment, psychiatric and medical comorbidities, and reduced quality of life[1]
- MDD is often chronic, with high risk of relapse and recurrence that may require long-term or lifetime treatment[2]
- Most patients fail to achieve remission (≈75%) or response (≈60%) with first-line antidepressant treatment (ADT) and required treatment switch or adjunctive treatment[3]
 - Inadequate ADT response is associated with increased hospitalization and suicide risk and greater impairments in functioning[4]
- Currently, the only FDA-approved adjunctive treatment options for MDD are atypical antipsychotics that have safety and tolerability concerns that impact short- and long-term medication adherence, including weight gain, cardiometabolic disturbances, and extrapyramidal symptoms (EPS)[5]
- Lumateperone is a mechanistically novel antipsychotic that is currently FDA-approved to treat schizophrenia and depressive episodes associated with bipolar I or bipolar II disorder as monotherapy and as adjunctive therapy with lithium or valproate[6]
 - Lumateperone is a simultaneous modulator of serotonin, dopamine, and glutamate neurotransmission[7]
 - Specifically, lumateperone is a potent serotonin 5-HT_{2A} receptor antagonist, a dopamine D_2 receptor presynaptic partial agonist and postsynaptic antagonist, a D_1 receptor-dependent indirect modulator of AMPA and NMDA currents, and a serotonin reuptake inhibitor[7]
 - This novel mechanism of action with multimodal effects may confer robust efficacy with improved safety and tolerability compared with current treatment options
- The efficacy and safety of lumateperone 42 mg adjunctive to ADT was recently demonstrated in 2 Phase 3 trials (Study 501 NCT04985942; Study 502 NCT05061706)[8,9]
- This Phase 3 open-label extension (OLE) trial, Study 503 (NCT05061719), examined the long-term safety and antidepressant effects of adjunctive lumateperone 42 mg in patients with MDD who had completed Study 501 or 502

METHODS

- Studies 501 and 502 enrolled patients aged 18-65 years, meeting DSM-5 criteria for MDD with inadequate response to 1-2 adequate courses of ADT in the current depressive episode and Montgomery-Åsberg Depression Rating Scale (MADRS) Total score ≥24 and Clinical Global Impression-Severity (CGI-S) score ≥4
- Patients who safely completed the 6-week double-blind treatment period could enroll in Study 503, which was a long-term OLE study to support FDA approval; all patients received open-label, oral, once-daily lumateperone 42 mg adjunctive to continued ADT
- During Study 503, lumateperone 42 mg was administered once daily in the evening for 26 weeks
- The primary endpoint was safety and tolerability of lumateperone 42 mg, measured by adverse events (AEs), EPS, suicidality, and changes in laboratory parameters, vital signs, physical examinations, and electrocardiogram (ECG) measures
 - EPS were assessed using a narrow standard MedDRA query for EPS-related treatment-emergent AEs (TEAEs) and the clinician-rated Abnormal Involuntary Movement Scale (AIMS), Barnes Akathisia Rating Scale (BARS), and Simpson-Angus Scale (SAS)
- The secondary endpoint was efficacy for depression symptoms, as measured by MADRS Total score and CGI-S score change from Study 501 or 502 baseline to Week 26 of open-label treatment

RESULTS

Patient Population

- Of the 809 patients enrolled in the OLE safety population, 684 patients (84.5%) completed the treatment period (**Table 1**)
 - The most common reasons for treatment discontinuation were adverse events (7.4%) and withdrawal of consent (5.1%)

Table 1. Patient Disposition of Safety Population

	Lumateperone 42 mg + ADT
Enrolled in OLE, n	812
Safety population, n	809
Discontinued treatment, n (%)[a]	125 (15.5)
Adverse event	60 (7.4)
Patient withdrew consent	41 (5.1)
Protocol violation	8 (1.0)
Lack of efficacy	7 (0.9)
Lost to follow-up	5 (0.6)
Other	4 (0.5)
Completed the OLE treatment period n (%)[a]	684 (84.5)

[a] Proportion of patients treated.
ADT, antidepressant therapy; OLE, open-label extension.

- Demographics and baseline characteristics were similar to patients in the 6-week double-blind placebo-controlled treatment periods (**Table 2**)
- The most common selective serotonin reuptake inhibitor was citalopram/escitalopram (30.4) and the most common serotonin-norepinephrine reuptake inhibitor was venlafaxine/desvenlafaxine (18.7)
- Mean MADRS and CGI-S scores at double-bind baseline indicated moderate-to-severe depression (**Table 2**)

Table 2. Baseline Demographics and Disease Characteristics in Safety Population

	Lumateperone 42 mg + ADT (N=809)
Age, mean (range), years	46.2 (18-66)
Sex, n (%)	
Female	549 (67.9)
Male	260 (32.1)
Race, n (%)	
White	703 (86.9)
Asian	61 (7.5)
Black	37 (4.6)
Other	8 (1.0)
Hispanic or Latino ethnicity, n (%)	86 (10.6)
No. of lifetime depressive episodes, mean (range)	3.8 (1-36)
No. of lifetime of treatment failures n (%)[a]	
1	596 (73.7)
2	213 (26.3)
Background ADT during the OLE	
SSRI	535 (66.1)
SNRI	224 (27.7)
Other (bupropion)	50 (6.2)
MADRS Total score	
At DB baseline, mean (SD)	30.7 (3.8)
At OLE baseline, mean (SD)	18.2 (8.6)
CGI-S score	
At DB baseline, mean (SD)	4.7 (0.6)
At OLE baseline, mean (SD)	3.4 (1.1)

[a] Includes the current episode. Treatment failure was defined as no remission with medications approved for the treatment of MDD at an adequate dose (per product label) and for an adequate duration of at least 6 weeks for monotherapy and 3 weeks for adjunctive therapy.
ADT, antidepressant therapy; CGI-S, Clinical Global Impression-Severity; DB, double-blind; MADRS, Montgomery-Åsberg Depression Rating Scale; MDD, major depressive disorder; OLE, open-label extension; SNRI, serotonin-norepinephrine reuptake inhibitor; SSRI, selective serotonin reuptake inhibitor.

Safety

Adverse Events

- Of 809 patients, 548 (67.7%) had ≥1 TEAE during the OLE (**Table 3**)
 - Most TEAEs (>98%) were mild or moderate in severity
 - Serious adverse events were 1.0%
- TEAEs that occurred in ≥5% of patients were headache, dizziness, dry mouth, nausea, somnolence, diarrhea, and nasopharyngitis
- AEs led to discontinuation in 7.4% of patients; only dizziness led to discontinuation in more than 1% of patients (1.1%)
- No suicidal behavior (per Columbia Suicide Severity Rating Scale) or suicidality serious AEs were reported in the study

Table 3. Adverse Events in Safety Population During OLE Period[a]

Event, n (%)	Lumateperone 42 mg + ADT (N=809)
≥1 TEAE	548 (67.7)
Drug-related TEAE	292 (36.1)
SAE	8 (1.0)
Discontinued treatment due to AE	60 (7.4)
Deaths	0
TEAEs occurring in ≥5% of patients	
Headache	134 (16.6)
Dizziness	86 (10.6)
Dry mouth	65 (8.0)
Nausea	62 (7.7)
Somnolence	58 (7.2)
Diarrhea	50 (6.2)
Nasopharyngitis	42 (5.2)

[a] An AE that occurred during the open-label treatment period was considered a TEAE if it started as a new event or if its severity increased during the OLE period; an AE that occurred more than 1 day after the date of the last dose of open-label lumateperone was not counted as a TEAE.
ADT, antidepressant therapy; AE, adverse event; OLE, open-label extension; SAE, serious adverse event; TEAE, treatment-emergent adverse event.

Body Morphology, Metabolic, Prolactin, and Vital Sign Assessments

- Changes in body morphology were small (**Table 4**)
- Potentially clinically significant weight increase or decrease (≥7% change from baseline) was low and similar
- There were minimal changes in cardiometabolic parameters (**Table 4**)
 - Mean changes in blood pressure, heart rate, and respiratory rate were minimal
- Mean changes in prolactin levels at end of treatment were low, similar to what was seen in double-blind studies, and not clinically relevant

Table 4. Mean Change in Body Morphology, Cardiometabolic Parameters, and Prolactin During OLE Period

	Lumateperone 42 mg + ADT (N=809)	
	Baseline Mean (SD)	**Mean Change From Baseline to EOT (SD)**
Weight, kg	78.96 (16.9)	−0.16 (3.72)
BMI, kg/m²	27.8 (5.06)	−0.05 (1.33)
Waist circumference, cm	92.6 (13.7)	−0.54 (5.50)
Cholesterol, mg/dL		
Total	199.7 (42.10)	−8.2 (32.30)
LDL	138.4 (41.24)	−9.6 (30.42)
HDL	56.7 (16.94)	0.1 (11.79)
Triglycerides, mg/dL	137.3 (81.66)	−0.2 (84.26)
Glucose, mg/dL	93.3 (14.75)	1.1 (15.56)
Insulin, μIU/L	14.51 (19.96)	−0.41 (22.41)
Hemoglobin A1c, %	5.6 (0.44)	0.0 (0.34)
Prolactin, ng/mL	10.07 (12.98)	1.13 (13.01)
PCS criterion	**n/N (%)**	
≥7% increase in weight	66/779 (8.5)	
≥7% decrease in weight	75/779 (9.6)	

ADT, antidepressant therapy; BMI, body mass index; EOT, end of treatment; HDL, high-density lipoprotein; LDL, low-density lipoprotein; PCS, potentially clinically significant.

Extrapyramidal Symptoms Assessments

- There were no notable changes in EPS as assessed by clinician-rated scales during the study (**Table 5**)
- EPS as defined by categorical shifts in the BARS or SAS scales were rare (**Table 5**)
- The frequency of EPS-related TEAEs was 3.8%

Table 5. Changes in EPS-Related Scales During OLE Period

	Lumateperone 42 mg + ADT	
	Baseline Mean (SD)	**Mean Change to EOT (SD)**
BARS Total score	0.1 (0.52)	−0.1 (0.53)
AIMS Total score	0.0 (0.37)	−0.0 (0.38)
SAS Total score	0.1 (0.36)	−0.0 (0.38)
EPS defined by categorical shifts	**n/N (%)[a]**	
Parkinsonism: Baseline SAS >3 during treatment	4/776 (0.5)	
Akathisia: Baseline BARS >2 during treatment	14/770 (1.8)	

[a] n = number of patients who met criteria at least once during the OLE period; N = the number of patients with available baseline assessments that did not meet the criteria and had at least one assessment during the OLE period.
ADT, antidepressant therapy; AIMS, Abnormal Involuntary Movement Scale; BARS; Barnes Akathisia Rating Scale; EOT, end of treatment; EPS, extrapyramidal symptoms; OLE, open-label extension; SAS, Simpson-Angus Scale.

Assessment of Depression Symptoms

- Improvement in depression symptoms continued throughout the OLE period, as measured by change in MADRS Total score (**Figure 1A**)
 - Similar improvement was seen in CGI-S score (data not shown)
- Most patients showed clinically meaningful improvements as assessed by MADRS response (79.5%) and remission (65.4%) criteria (**Figure 1B**)

Figure 1. Assessment of Depression Symptoms: Mean Change From Baseline in MADRS Total Score (A) and MADRS Response and Remission Rates (B)

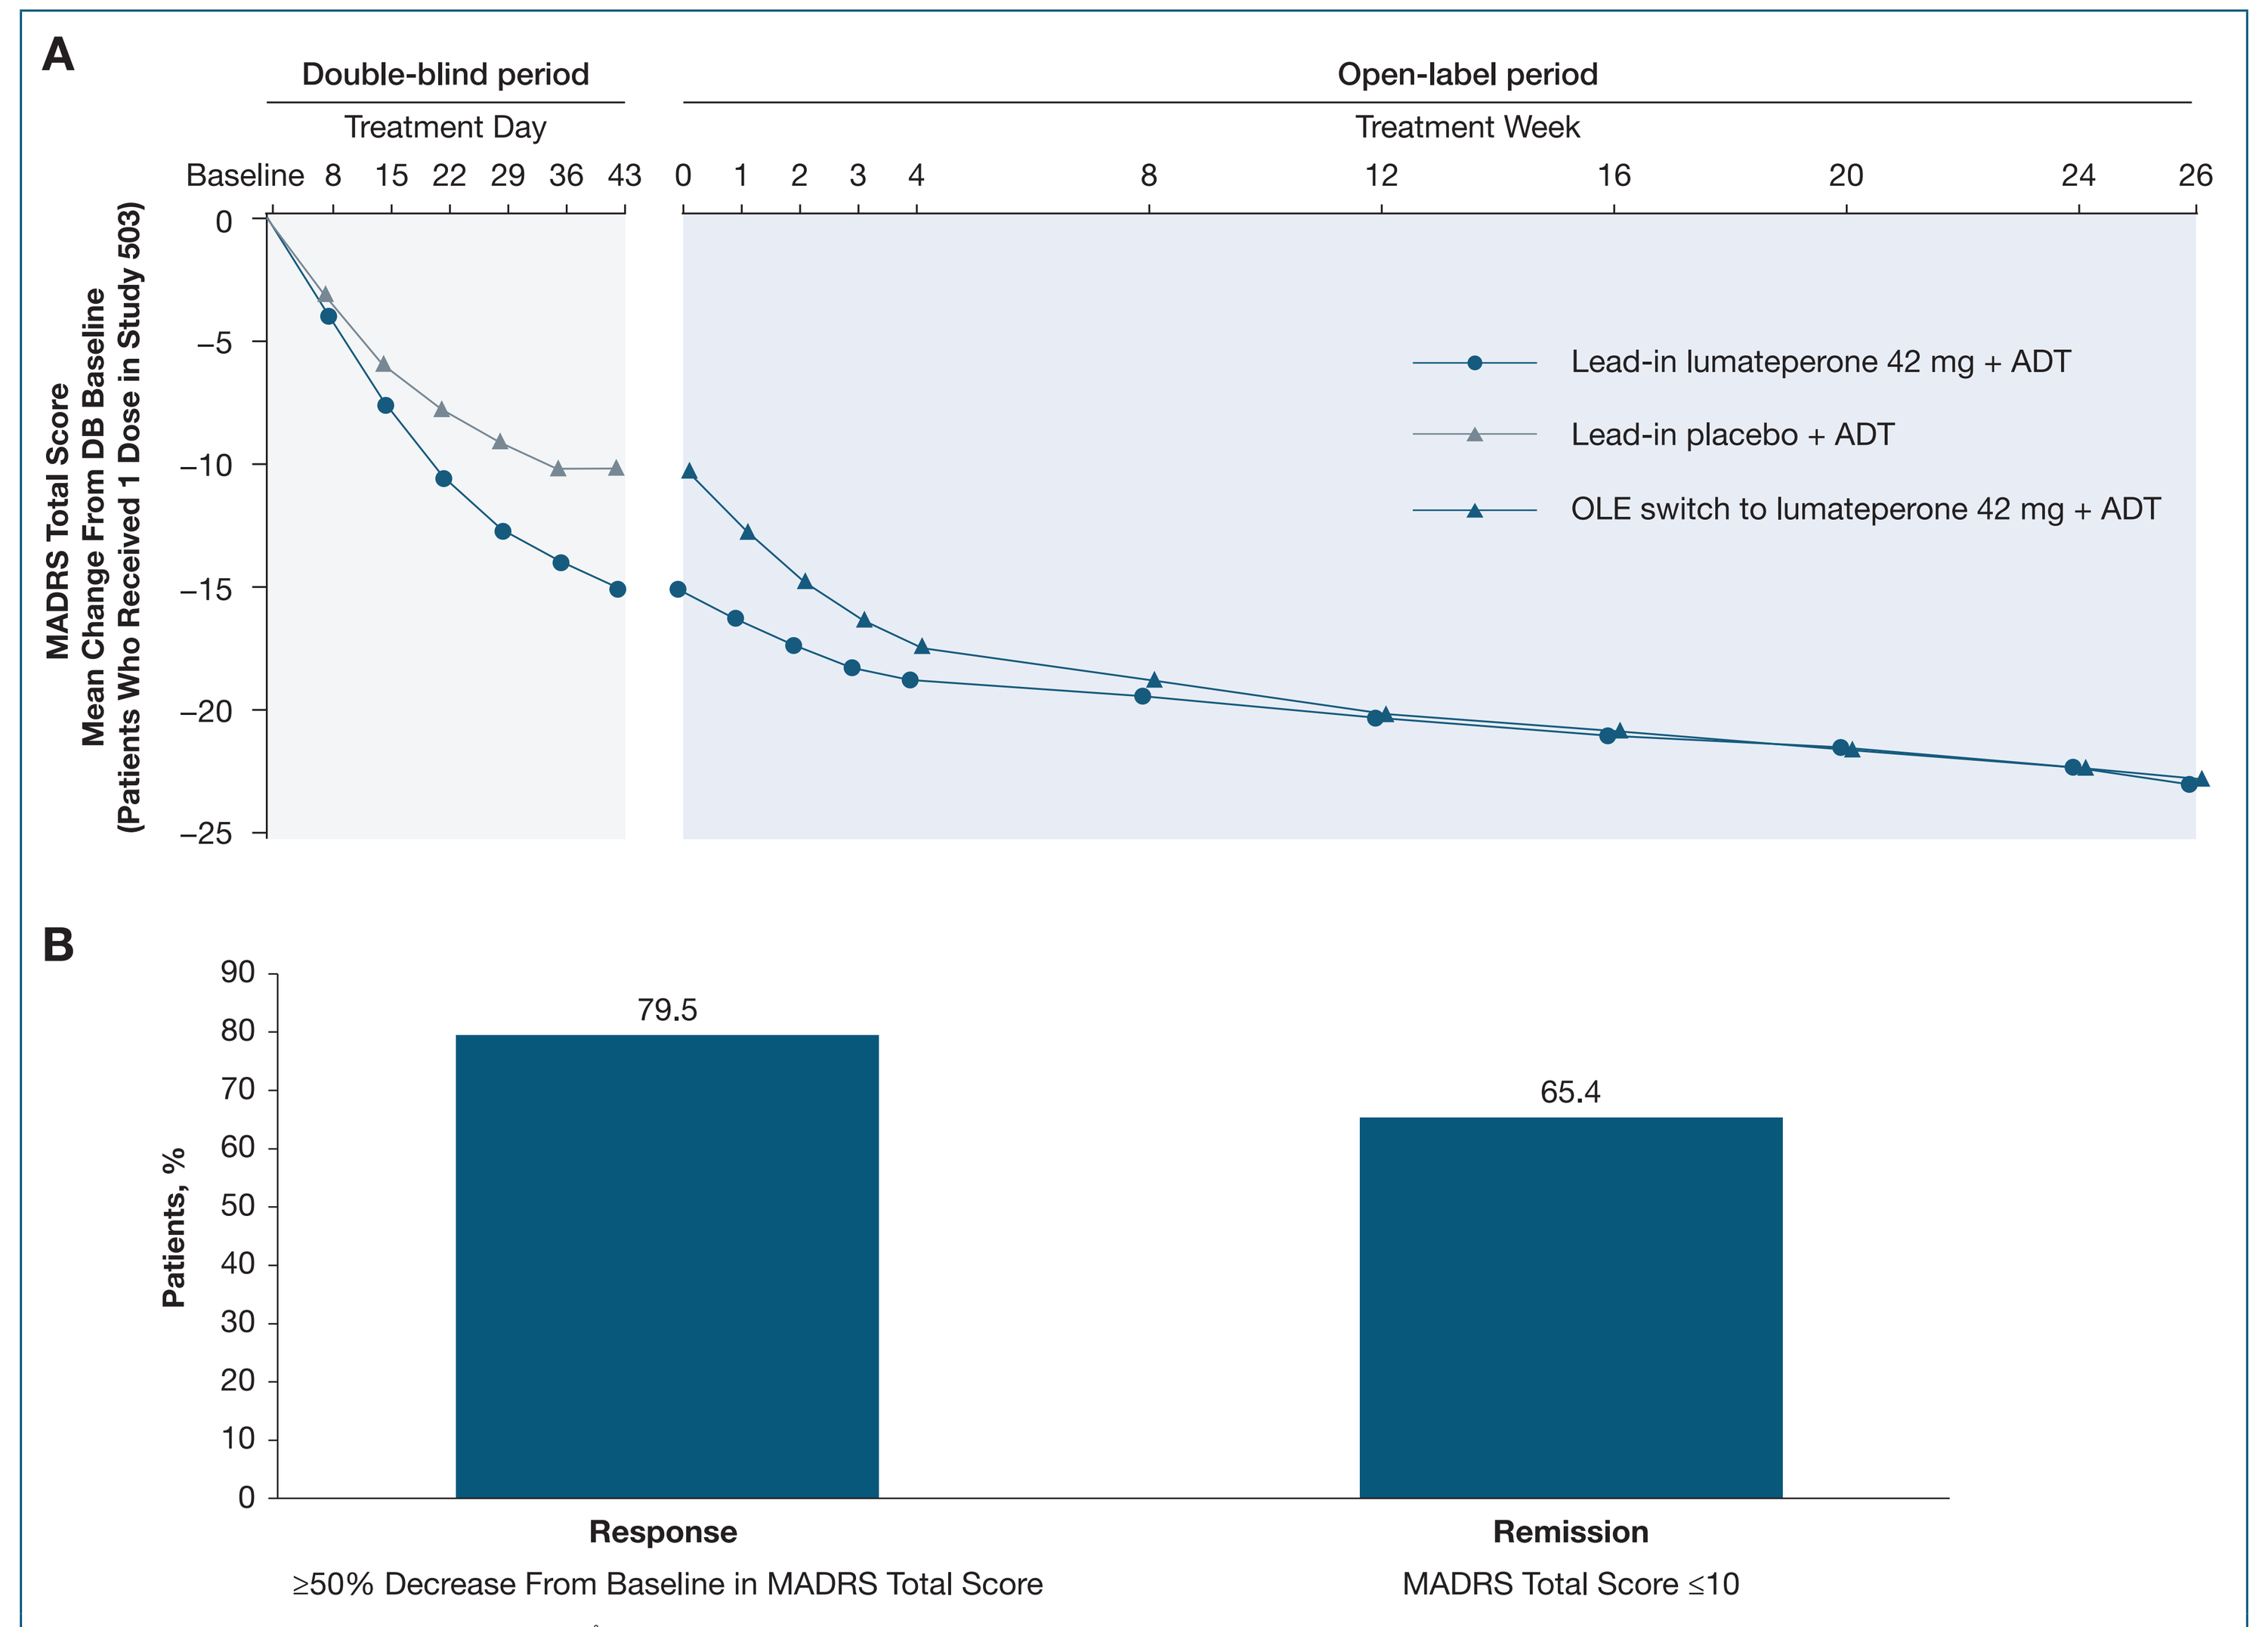
A
Double-blind period
Treatment Day
Open-label period
Treatment Week
Baseline 8 15 22 29 36 43
0 1 2 3 4 8 12 16 20 24 26
MADRS Total Score Mean Change From DB Baseline (Patients Who Received 1 Dose in Study 503)
0
−5
−10
−15
−20
−25
Lead-in lumateperone 42 mg + ADT
Lead-in placebo + ADT
OLE switch to lumateperone 42 mg + ADT
B
Patients, %
90
80
70
60
50
40
30
20
10
0
79.5
65.4
Response
≥50% Decrease From Baseline in MADRS Total Score
Remission
MADRS Total Score ≤10

ADT, antidepressant therapy; DB, double-blind; MADRS, Montgomery-Åsberg Depression Rating Scale; OLE, open-label extension.

CONCLUSIONS

- **Lumateperone 42 mg adjunctive to ADT was generally safe and well tolerated in long-term treatment in patients with MDD; there were no new safety findings, and AEs and safety parameters were consistent with the short-term 501 and 502 studies**
- **Over 26 weeks of treatment, lumateperone 42 mg adjunctive to ADT was associated with low risk of weight gain, cardiometabolic effects, and EPS**
- **In patients treated long-term with lumateperone 42 mg adjunctive to ADT, efficacy was maintained, and symptoms of depression improved throughout the study**
- **These results support the long-term safety and effectiveness of lumateperone 42 mg adjunctive to ADT in patients with MDD and inadequate ADT response**

REFERENCES

1. Proudman D, et al. *Pharmacoeconomics.* 2021;39(6):619-625.
2. Lam RW, et al. *Can J Psychiatry.* 2024;69(9):641-687.
3. Pigott HE, et al. *BMJ Open.* 2023;13(7):e063095.
4. Knoth RL, et al. *Am J Manag Care.* 2010;16(8):e188-e196.
5. Spielmans GI, et al. *PLoS Med.* 2013;10(3):e100140310.
6. Caplyta. Prescribing information. Intra-Cellular Therapies, Inc.;2023.
7. Titulaer J, et al. *Eur Neuropsychopharmacol.* 2022;62:22-35.
8. Durgam S, et al. *J Clin Psychiatry.* 2025;86(4):25m15848.
9. Durgam S, et al. *Am J Psychiatry.* In Press.

DISCLOSURES AND ACKNOWLEDGMENTS

WR Earley, S Durgam, C Chen, and F Brennan are full-time employees of Intra-Cellular Therapies, a Johnson & Johnson Company. SG Kozauer is a former employee of Intra-Cellular Therapies, a Johnson & Johnson Company.

AJ Cutler has served as a consultant/on an advisory board for: AbbVie, Acadia, Alfasigma, Alkermes, Anavex Life Sciences, Autobahn Therapeutics, Axsome, Biogen, Biohaven, Boehringer Ingelheim, Brii Biosciences, Bristol Myers Squibb, Cerevel, Corium, Delpor, Evolution Research Group, 4M Therapeutics, Intra-Cellular Therapies, J&J Innovative Medicine, Jazz Pharma, Karuna, LivoNova, Lundbeck, Luye Pharma, MapLight Therapeutics, MedAvante-ProPhase, Mentavi, Neumora, Neurocrine, Neuroscience Education Institute, NeuroSigma, Noven, Otsuka, PaxMedica, Relmada, Sage Therapeutics, Supernus, Teva, Thynk, Tris Pharma, Vanda Pharmaceuticals, and VistaGen; Served on a speaker's bureau for: AbbVie, Alfasigma, Alkermes, Axsome, Bristol Myers Squibb, Corium, Intra-Cellular Therapies, J&J, Lundbeck, Neurocrine, Noven, Otsuka, Supernus, Teva, Tris Pharma, and Vanda Pharmaceuticals; Owns stock options with: 4M Therapeutics; Served on a data safety monitoring board for: Alar Pharma, COMPASS Pathways, Freedom Biosciences, and Pain Therapeutics.

Key contributors: All authors: data analysis/interpretation and writing/critical review. Additionally, WR Earley, S Durgam, SG Kozauer, and C Chen participated in study design/conduct.

The authors thank all study investigators, research staff, and patients for their participation. Medical writing support was provided by Adam Ruth, PhD, of Nucleus Global, an Inizio company, funded by Intra-Cellular Therapies, a Johnson & Johnson Company.

Scan the QR code for the full digital poster. The QR code is intended to provide scientific information for individual reference, and the information should not be altered or reproduced in any way.

Presented at the Neuroscience Education Institute Fall Congress, November 6-9, 2025, Colorado Springs, CO

Email questions to Francis Brennan at fbrennan@itci-inc.com