Participants With Active, Erosive Psoriatic Arthritis: Post hoc Analysis of the APEX Study at Week 24

Christopher T. Ritchlin,[1] Proton Rahman,[2] Laura C. Coates,[3] Alexa P. Kollmeier,[4] Yusang Jiang,[5] Molly Caselton,[6] Soumya D. Chakravarty,[6,7] Elizabeth Adamson,[6] Karissa Lozenski,[6] Arthur Kavanaugh,[8] Philip J. Mease[9,10]

[1]Department of Medicine, Allergy/Immunology and Rheumatology, University of Rochester Medical Center, Rochester, NY, USA; [2]Faculty of Medicine, Division of Rheumatology, Memorial University of Newfoundland, St. Johns, NL, Canada; [3]Nuffield Department of Orthopaedics, Rheumatology and Musculoskeletal Sciences, University of Oxford, Botnar Research Centre, Oxford, UK; [4]Johnson & Johnson, San Diego, CA, USA; [5]Johnson & Johnson, Spring House, PA, USA; [6]Johnson & Johnson, Horsham, PA, USA; [7]Drexel University College of Medicine, Philadelphia, PA, USA; [8]Center for Innovative Therapy, Division of Rheumatology, Allergy, Immunology, University of California San Diego, La Jolla, CA, USA; [9]Rheumatology Research, Providence Swedish Medical Center, Seattle, WA, USA; [10]University of Washington School of Medicine, Seattle, WA, USA

Scan the QR code.
The QR code is intended to provide scientific information for individual reference, and the information should not be altered or reproduced in any way.

Key Takeaways

- In the APEX population of pts with active and erosive PsA, GUS treatment significantly inhibited structural damage progression vs PBO at W24[2]
- Among pts receiving GUS every 4 weeks and every 8 weeks, achievement of clinical response was associated with less structural damage progression at W24 across PsA domains
- Additional analyses beyond W24 will determine the maintenance of structural damage inhibition among clinical responders and continue assessment of clinical nonresponders

Background

Guselkumab (GUS) inhibition of structural damage progression in psoriatic arthritis (PsA)

- Uncontrolled inflammation in PsA increases the risk of structural joint damage, which is associated with long-term disability and greater impairments in physical function and health-related quality of life
- GUS is a fully human, dual-acting,[1] monoclonal antibody that selectively inhibits the interleukin (IL)-23p19 subunit
- In the phase 3b APEX study, GUS demonstrated efficacy across PsA domains and significantly inhibited structural damage progression compared with placebo (PBO) in participants (pts) with active and erosive PsA at Week (W) 24[2]

Objective

Evaluate the relationship between clinical response status and structural damage progression at W24 among GUS-randomized pts in APEX

Methods

Inclusion Criteria

- ✓ Biologic-naive
- ✓ ≥18 years
- ✓ Active PsA ≥6 months (despite prior csDMARDs, apremilast, NSAIDs); CASPAR criteria met
- ✓ ≥3 SJC; ≥3 TJC; CRP ≥0.3 mg/dL
- ✓ **≥2 joints with erosions** on baseline radiographs of hands/foots (centrally read)
- ✓ Active plaque PsO (≥1 PsO plaque ≥2 cm and/or nail PsO)

APEX Study Design

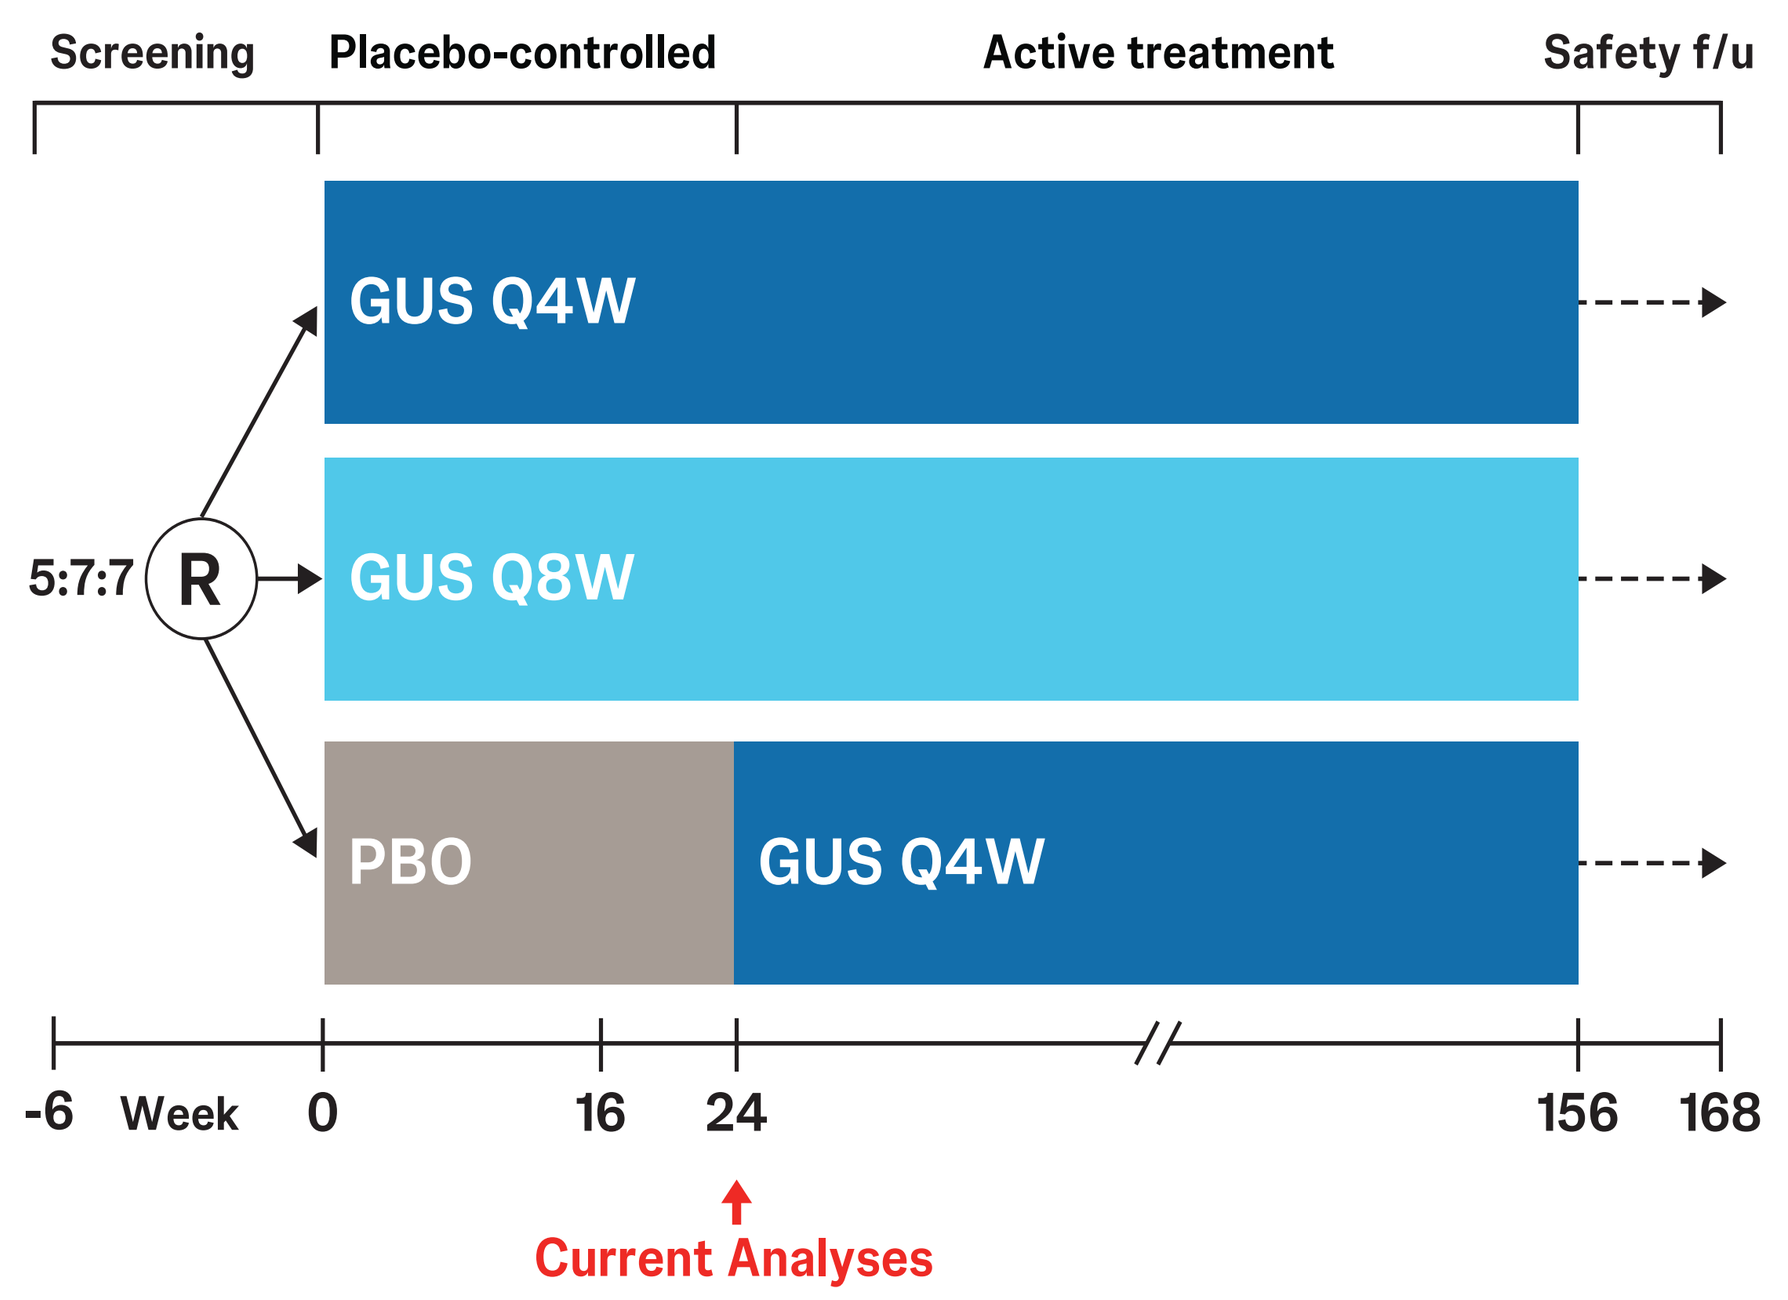
Screening
Placebo-controlled
Active treatment
Safety f/u
5:7:7
R
GUS Q4W
GUS Q8W
PBO
GUS Q4W
-6
Week
0
16
24
156
168
Current Analyses

Endpoints assessed at W24

LSM change from baseline in PsA-modified vdH-S score by clinical response status at W24, as assessed by:

- ACR20/50/70
- DAPSA LDA ≤14/REM ≤4[3]
- cDAPSA LDA ≤13/REM ≤4[3,4]
- MDA[5]
- HAQ-DI ≤0.5
- Pt-Reported Pain ≤15 mm on VAS

Statistical Analyses

- Pts were considered clinical nonresponders if they initiated/increased oral corticosteroids, csDMARDs, initiated protocol-prohibited PsA therapies, or discontinued study agent for any reason other than ND/MD
 - Data affected by ND/MD were not used
- For change in vdH-S scores, data affected by ND/MD were not used; missing data were imputed using MI

ACR20/50/70=≥20%/≥50%/≥70% improvement in the American College of Rheumatology criteria, CASPAR=ClASsification criteria for Psoriatic Arthritis, cDAPSA=clinical Disease Activity Index for Psoriatic Arthritis, CRP=C-reactive protein, csDMARDs=conventional synthetic disease modifying antirheumatic drugs, DAPSA=Disease Activity Index for Psoriatic Arthritis, f/u=follow-up, GUS=guselkumab, HAQ-DI=Health Assessment Questionnaire-Disability Index, LDA=low disease activity, LSM=least squares mean, MD=major disruption (Ukraine/Russia Crisis), MDA=minimal disease activity, MI=multiple imputation, ND=natural disaster (COVID-19 site access restrictions), NSAIDs=nonsteroidal anti-inflammatory drug, PBO=placebo, PsA=psoriatic arthritis, PsO=psoriasis, Pts=participants, Q4W=every 4 weeks, Q8W=every 8 weeks, R=randomization, REM=remission, SJC=swollen joint count, TJC=tender joint count, VAS=visual analog scale, vdH-S=van der Heijde-Sharp, W=week.

Results

Baseline characteristics were well balanced across treatment groups

	GUS Q4W (N=273)	GUS Q8W (N=371)	PBO (N=376)	Total (N=1020)
Demographics				
Age, years	52.2 (13.2)	53.2 (12.9)	53.5 (13.0)	53.0 (13.0)
Male	55%	54%	57%	55%
Weight, kg	85.6 (20.1)	83.2 (17.4)	83.1 (18.2)	83.8 (18.5)
BMI, kg/m²	29.4 (6.0)	29.0 (5.6)	28.9 (5.7)	29.1 (5.7)
PsA Characteristics				
PsA Disease Duration, years	7.5 (7.1)	7.2 (7.6)	7.2 (6.9)	7.3 (7.2)
SJC [0–66]	11.6 (9.4)	12.1 (8.5)	11.8 (8.9)	11.9 (8.9)
TJC [0–68]	21.2 (14.6)	20.6 (13.4)	20.5 (13.9)	20.7 (13.9)
Pt-reported pain (VAS; 0–100 mm)	59.2 (22.2)	59.3 (21.0)	59.4 (20.6)	59.3 (21.1)
HAQ-DI [0–3]	1.2 (0.7)	1.2 (0.6)	1.2 (0.7)	1.2 (0.7)
DAPSA	46.4 (24.3)	46.1 (22.1)	46.0 (22.8)	46.1 (22.9)
cDAPSA	44.7 (23.5)	44.6 (21.6)	44.3 (22.3)	44.5 (22.4)
CRP, mg/dL	1.7 (2.9)	1.5 (2.0)	1.7 (2.5)	1.6 (2.5)
Enthesitis / Dactylitis	58% / 44%	59% / 39%	59% / 45%	58% / 43%
Mean LEI [1–6]/**DSS** [1–60]	3.2 / 10.8	3.0 / 11.0	3.0 / 10.2	3.1 / 10.6
Radiographic Characteristics				
PsA-modified vdH-S score [0–528]	27.7 (47.6)	26.7 (43.4)	26.8 (42.2)	27.0 (44.1)
Erosion score [0–320]	13.7 (24.3)	13.4 (21.9)	13.4 (20.7)	13.5 (22.1)
JSN score [0–208]	14.0 (24.2)	13.3 (22.8)	13.4 (22.4)	13.5 (23.0)

Values are reported as mean (SD) for pts with nonmissing data unless otherwise noted. BMI=body mass index, cDAPSA=clinical Disease Activity Index for Psoriatic Arthritis, CRP=C-reactive protein, DAPSA=Disease Activity Index for Psoriatic Arthritis, DSS=Dactylitis Severity Score, GUS=guselkumab, HAQ-DI=Health Assessment Questionnaire-Disability Index, JSN=joint space narrowing, LEI=Leeds Enthesitis Index, PBO=placebo, PsA=psoriatic arthritis, Pts=participants, Q4W=every 4 weeks, Q8W=every 8 weeks, SD=standard deviation, SJC=swollen joint count, TJC=tender joint count, VAS=visual analog scale, vdH-S=van der Heijde-Sharp.

At W24, GUS-randomized pts who achieved response in joint efficacy measures had numerically lower levels of structural damage progression

Joint efficacy responses

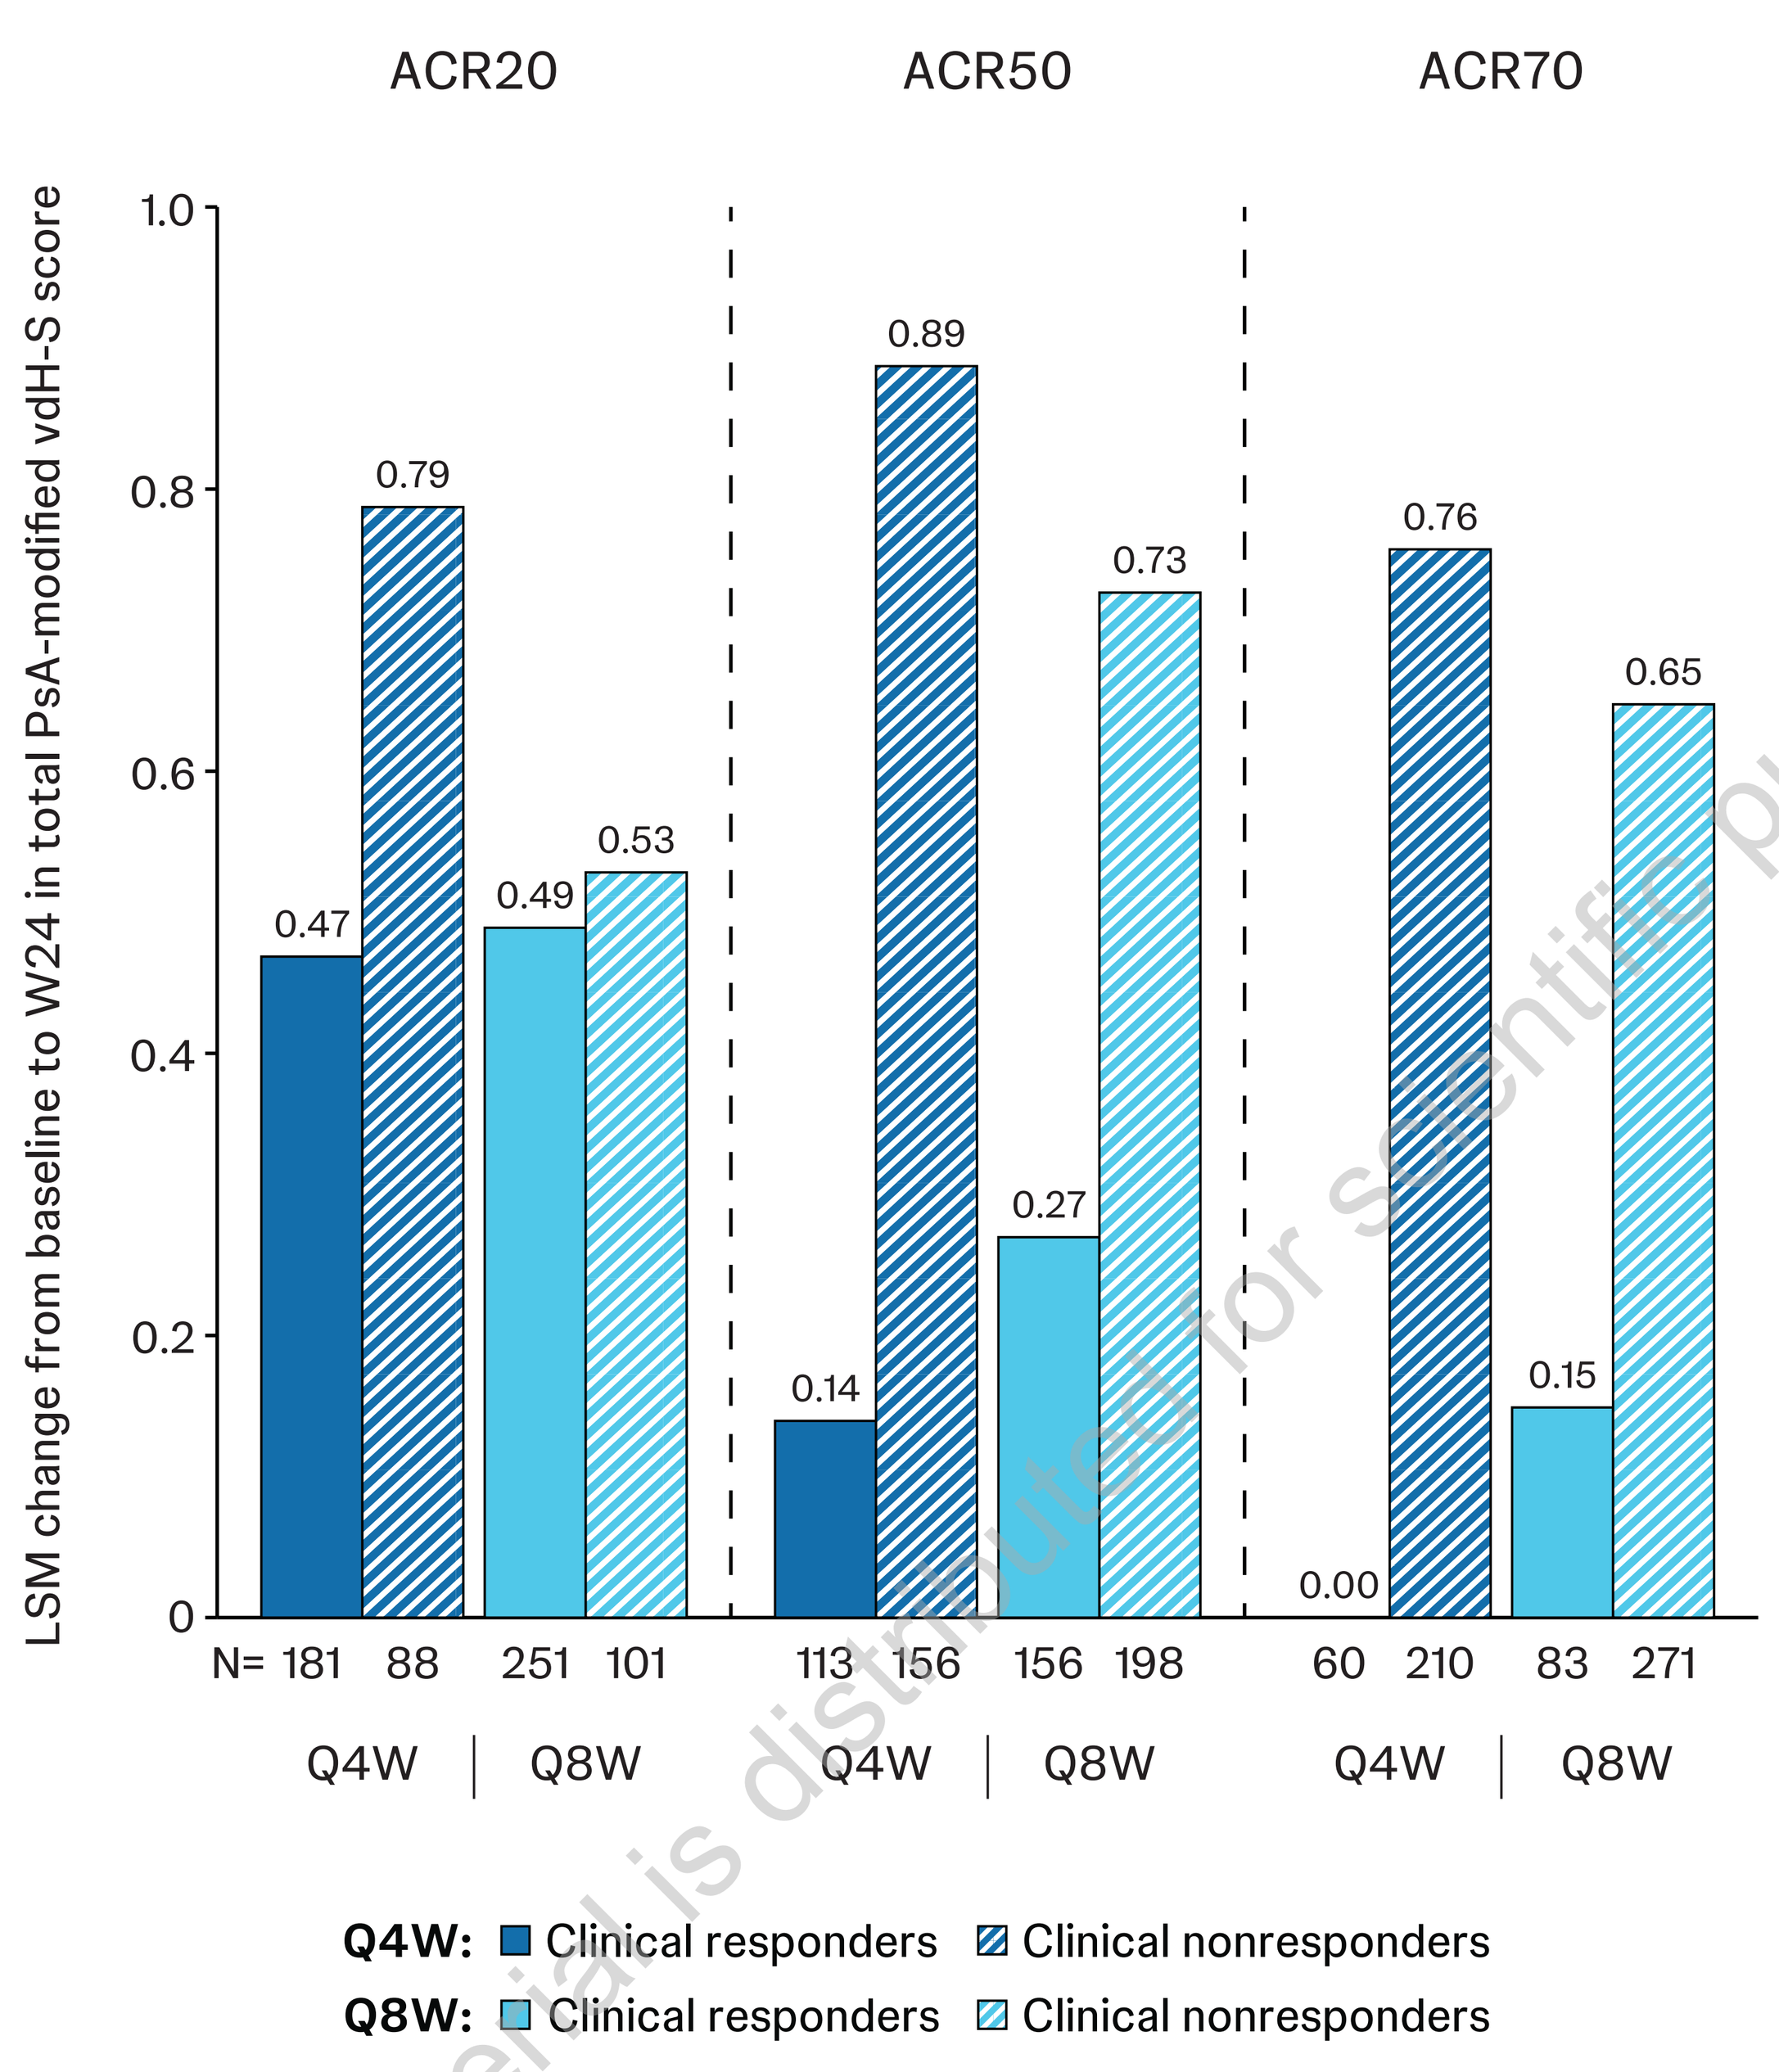
ACR20
ACR50
ACR70
LSM change from baseline to W24 in total PsA-modified vdH-S score
1.0
0.8
0.6
0.4
0.2
0
0.47
0.79
0.49
0.53
0.14
0.89
0.27
0.73
0.00
0.76
0.15
0.65
N= 181 88 251 101
113 156 156 198
60 210 83 271
Q4W | Q8W
Q4W | Q8W
Q4W | Q8W
Q4W: Clinical responders Clinical nonresponders
Q8W: Clinical responders Clinical nonresponders

ACR20/50/70=≥20%/≥50%/≥70% improvement in the American College of Rheumatology criteria, LSM=least squares mean, PsA=psoriatic arthritis, Q4W=every 4 weeks, Q8W=every 8 weeks, vdH-S=van der Heijde-Sharp, W=week.

At W24, GUS-randomized pts who achieved low or minimal disease activity by composite indices had numerically lower levels of structural damage progression

Composite disease outcome measures

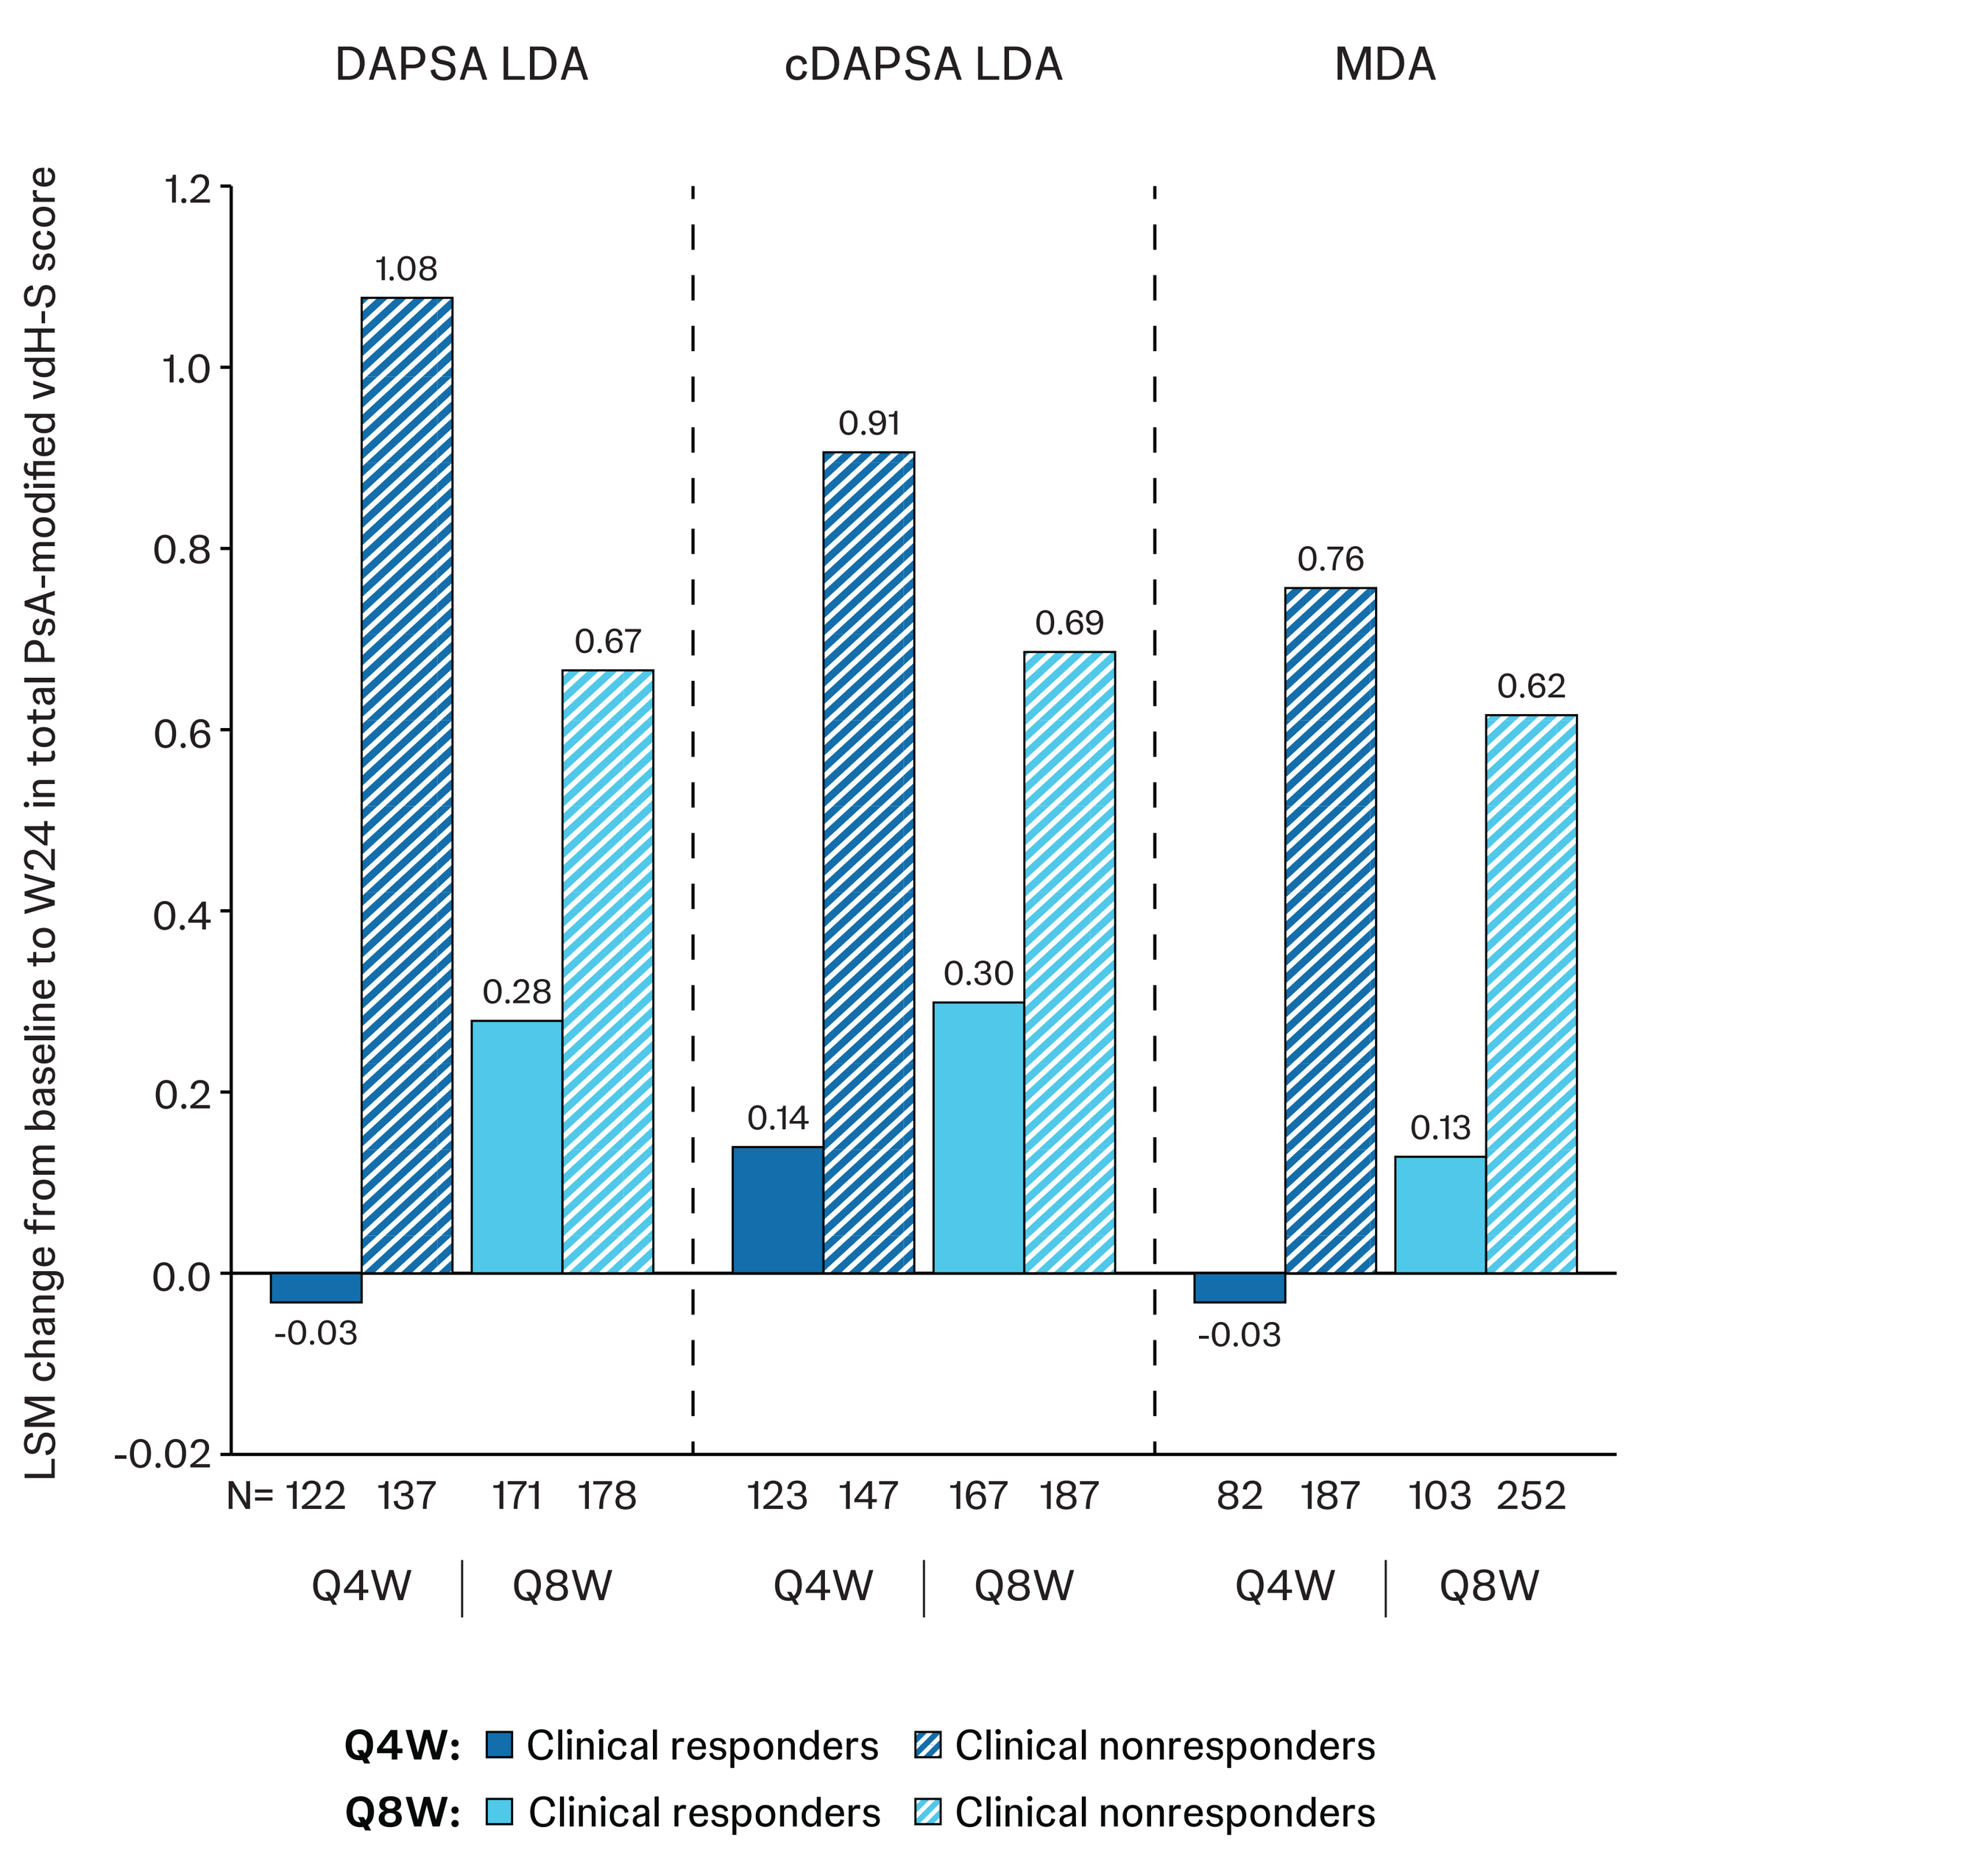
DAPSA LDA
cDAPSA LDA
MDA
LSM change from baseline to W24 in total PsA-modified vdH-S score
1.2
1.0
0.8
0.6
0.4
0.2
0.0
-0.02
-0.03
1.08
0.28
0.67
0.14
0.91
0.30
0.69
-0.03
0.76
0.13
0.62
N= 122 137 171 178
123 147 167 187
82 187 103 252
Q4W | Q8W
Q4W | Q8W
Q4W | Q8W
Q4W: Clinical responders Clinical nonresponders
Q8W: Clinical responders Clinical nonresponders

cDAPSA=clinical Disease Activity Index for Psoriatic Arthritis, DAPSA=Disease Activity Index for Psoriatic Arthritis, LDA=low disease activity, LSM=least squares mean, MDA=minimal disease activity, PsA=psoriatic arthritis, Q4W=every 4 weeks, Q8W=every 8 weeks, vdH-S=van der Heijde-Sharp, W=week.

At Week 24, GUS-randomized pts who achieved normalized physical function and minimal pain had numerically lower levels of structural damage progression

Patient-reported outcomes

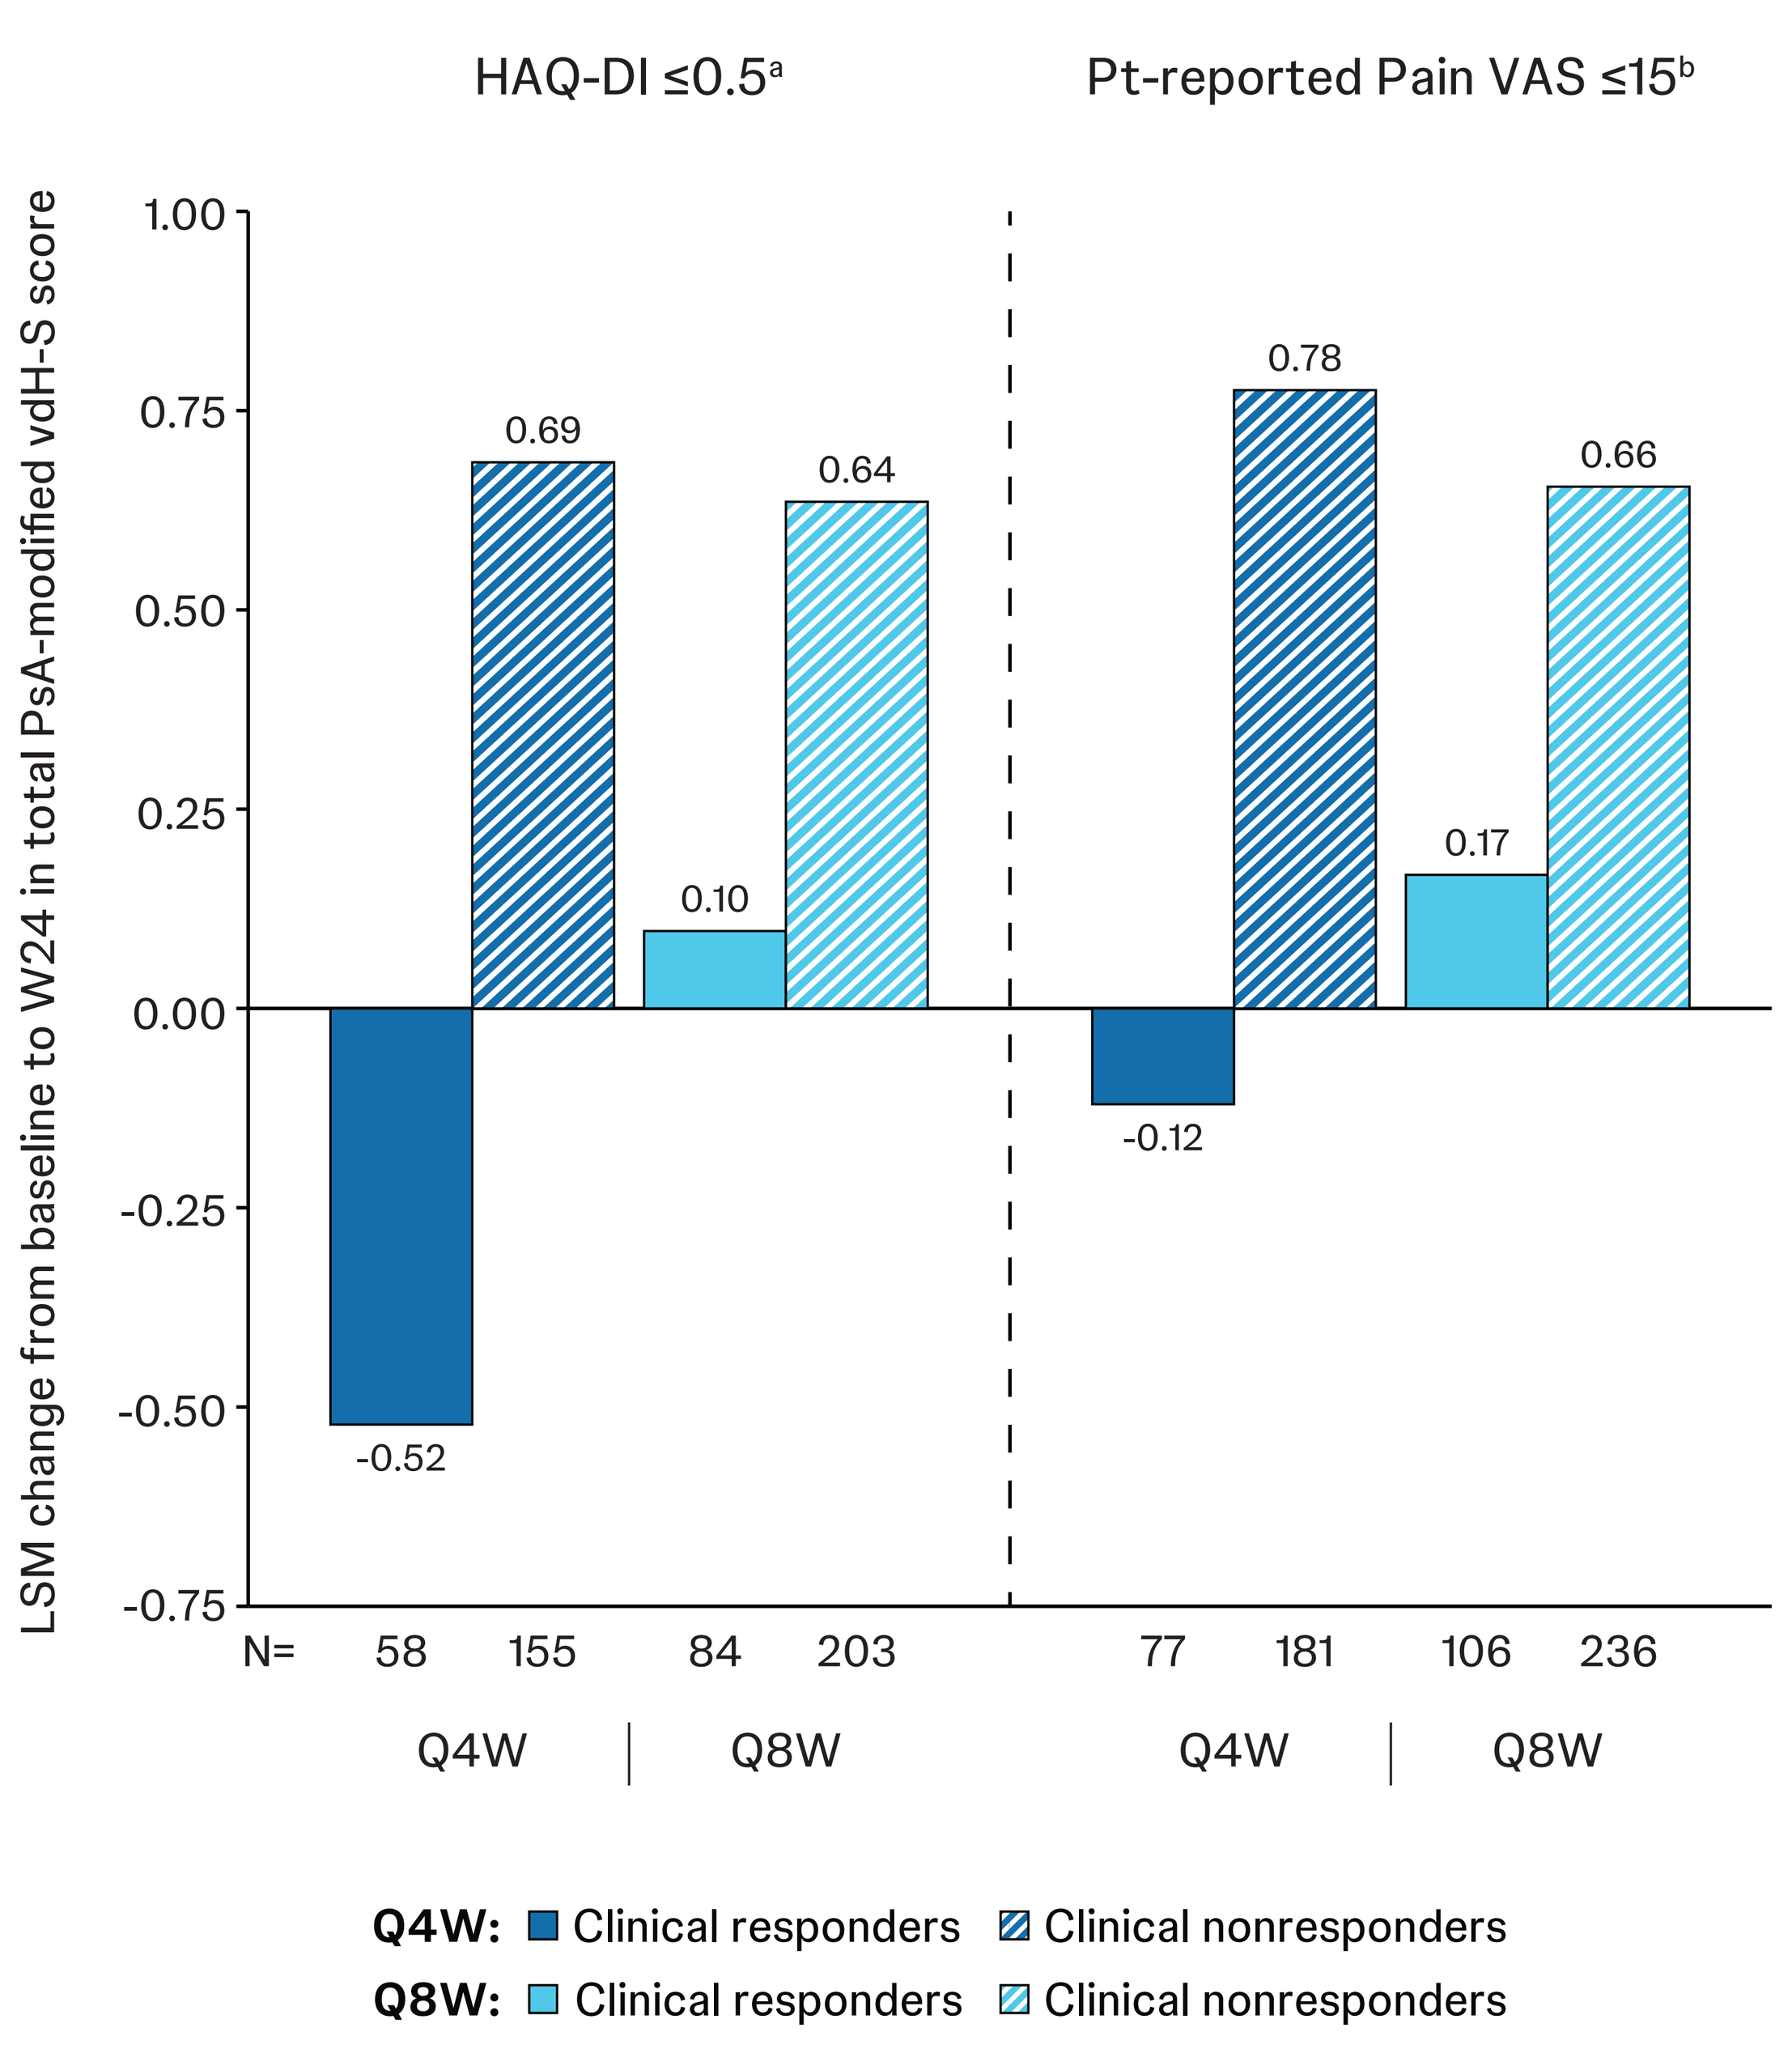
HAQ-DI ≤0.5[a]
Pt-reported Pain VAS ≤15[b]
LSM change from baseline to W24 in total PsA-modified vdH-S score
1.00
0.75
0.50
0.25
0.00
-0.25
-0.50
-0.75
-0.52
0.69
0.10
0.64
-0.12
0.78
0.17
0.66
N= 58 155 84 203
77 181 106 236
Q4W | Q8W
Q4W | Q8W
Q4W: Clinical responders Clinical nonresponders
Q8W: Clinical responders Clinical nonresponders

[a]Among pts who had HAQ-DI score >0.5 at baseline. [b]Among pts who had pt-reported pain VAS >15 at baseline. HAQ-DI=Health Assessment Questionnaire-Disability Index, LSM=least squares mean, PsA=psoriatic arthritis, Pts=participants, Q4W=every 4 weeks, Q8W=every 8 weeks, VAS=visual analog scale, vdH-S=van der Heijde-Sharp, W=week.

PRESENTED BY: Molly Caselton at the Congress of Clinical Rheumatology-West (CCR-West), September 17–20, 2026; Huntington Beach, California, USA. **REFERENCES: 1.** Sachen KL, et al. *Front Immunol.* 2025;16:1532852. **2.** Mease PJ. *Ann Rheum Dis.* 2025;84:1983–1984. **3.** Schoels M. *Ann Rheum Dis.* 2016;75:811–818. **4.** Helliwell PS. *Arthritis Care Res.* 2014;66:749–756. **5.** Coates LC. *J Rheumatol.* 2016;43:371–375. **ACKNOWLEDGEMENTS:** Medical writing support was provided by Kelly Koch, PharmD of Johnson & Johnson, under the direction of the authors in accordance with Good Publication Practice guidelines (Ann Intern Med. 2022;175:1298–1304). Layout design and formatting for this encore presentation was provided by Sandeep Chavan (SIRO Medical Writing Pvt. Ltd., India). This study and presentation were sponsored by Johnson & Johnson. **DISCLOSURES: CTR c**onsultant for AbbVie, Amgen, Eli Lilly, Gilead, Johnson & Johnson, Novartis, Pfizer, and UCB; received grant/research support: AbbVie, Amgen, and UCB. **PR c**onsultant for AbbVie, Amgen, Bristol Myers Squibb, Celgene, Eli Lilly, Johnson & Johnson, Merck, Novartis, Pfizer, and UCB; received grant/research support from Johnson & Johnson and Novartis; received meeting attendance/travel support from Johnson & Johnson. **LCC:** Consultant for AbbVie, Amgen, Bristol Myers Squibb, Celgene, Eli Lilly, Galapagos, Gilead, Johnson & Johnson, Moonlake, Novartis, Pfizer, and UCB; received grants/research support from AbbVie, Amgen, Celgene, Eli Lilly, Johnson & Johnson, Novartis, Pfizer and UCB; received manuscripts support from the National Institute for Health Research (NIHR) Oxford Biomedical Research Centre (BRC). The views expressed are those of the author(s) and not necessarily those of the NHS, the NIHR, or the Department of Health; is a paid speaker for AbbVie, Amgen, Biogen, Celgene, Eli Lilly, Galapagos, Gilead, GlaxoSmithKline, Johnson & Johnson, Medac, Novartis, Pfizer, and UCB. **APK, MC, SDC, EA,** and **KL** employee of Johnson & Johnson and may own stock/stock options in Johnson & Johnson. **YJ** employee of Cytel providing statistical support (funded by Johnson & Johnson). **AK c**onsultant for AbbVie, Amgen, Bristol Myers Squibb, Eli Lilly, Johnson & Johnson, Moonlake, Novartis, Pfizer, and UCB. **PJM** consultant for AbbVie, Amgen, Bristol Myers Squibb, Century, Cullinan, Eli Lilly, Inmagene, Johnson & Johnson, Merck, Moonlake, Novartis, Pfizer, Spyre, SUN Pharma, Takeda, and UCB; received grant/research support from AbbVie, Amgen, Bristol Myers Squibb, Eli Lilly, Johnson & Johnson, Moonlake, Novartis, Sana, Takeda, and UCB; is a paid speaker for AbbVie, Amgen, Eli Lilly, Johnson & Johnson, Novartis, and UCB. **PREVIOUSLY PRESNTED AT:** The European Alliance of Associations for Rheumatology (EULAR) European Congress; June 3–6, 2026; London, England, UK.

This material is distributed for scientific purposes by Johnson & Johnson Innovative Medicine and is not for promotional use.