Pregnancy Outcomes in Maternal Exposure to Guselkumab: Review of Cases Reported to the Company's Global Safety Database

Uma Mahadevan,[1] Millie Long,[2] Mette Julsgaard,[3] Connie Lin,[4] Anja Geldhof,[5] Mauricio Rosas Ballina,[6] Hewei Li,[7] Allison Hester,[4] Javier P. Gisbert,[8] María Chaparro[8]

[1]University of California, San Francisco, California, United States; [2]University of North Carolina, Chapel Hill, North Carolina, United States; [3]Department of Hepatology and Gastroenterology, Aarhus University Hospital, Aarhus, Denmark; [4]Johnson & Johnson, Horsham, Pennsylvania, United States; [5]Johnson & Johnson, Diegem, Belgium; [6]Actelion Research & Development, Basel, Switzerland; [7]Johnson & Johnson, Titusville, New Jersey, United States; [8]Hospital Universitario de La Princesa, Instituto de Investigación Sanitaria Princesa (IIS-Princesa), Universidad Autónoma de Madrid (UAM), and Centro de Investigación Biomédica en Red de Enfermedades Hepáticas y Digestivas (CIBEREHD), Madrid, Spain

Scan the QR code.
The QR code is intended to provide scientific information for individual reference, and the information should not be altered or reproduced in any way.

This material is distributed for scientific purposes by Johnson & Johnson Innovative Medicine and is not for promotional use.

Key Takeaways

- **These findings suggest no apparent impact of GUS on pregnancy outcomes; however, they should be interpreted cautiously given data limitations**
- **Further studies are warranted to confirm these observations and to better characterize the safety profile of GUS exposure during pregnancy**

The limitations of these findings include, but are not limited to, the lack of study control, a small sample size of pregnancies with known outcomes, and reported data with varying degrees of missing information.

Background

- The Global Consensus Consortium statement on the management of pregnancy in inflammatory bowel disease suggests women can continue treatment with interleukin (IL)-23 inhibitors throughout pregnancy[1]
- Guselkumab (GUS) is a dual-acting selective p19 subunit-targeted IL-23 inhibitor that blocks IL-23 signaling and binds to CD64, a receptor on immune cells that produce IL-23[2]
- GUS is approved to treat moderate-to-severe psoriasis (PsO), active psoriatic arthritis (PsA), and moderately to severely active ulcerative colitis (UC) and Crohn's disease (CD)[3]

Objectives

- **In this analysis, we report data on pregnancy cases with known outcomes in women exposed to GUS during pregnancy**

Methods

- **Pregnancy cases were reported to the Company's Global Safety Database through 12 July 2025**
 - Cases with known outcomes were reported from clinical studies and spontaneous sources
- **Data were summarized descriptively for pregnancies reported prospectively and retrospectively**
- **GUS therapeutic indications in pregnancy cases were:**
 - Psoriatic disease
 - CD
 - UC
 - Other/not reported
- **Maternal GUS exposure was categorized as follows:**
 - Before conception (within 3 months prior to conception)
 - During the first trimester (T1)
 - After the first trimester only (T2, T3)
 - Throughout pregnancy
 - Not reported
- **Pregnancy outcomes were classified as:**
 - Live births with or without congenital anomalies
 - Spontaneous abortions
 - Elective terminations with or without fetal defects or unknown
 - Ectopic pregnancies
 - Stillbirths with or without fetal defects
 - Unspecified abortions with or without fetal defects or unknown

Results

A total of 400 pregnancy events with known outcomes occurred among 396 women

- Maternal age was reported for 264/396 (67%) women, and the mean maternal age was 32 years

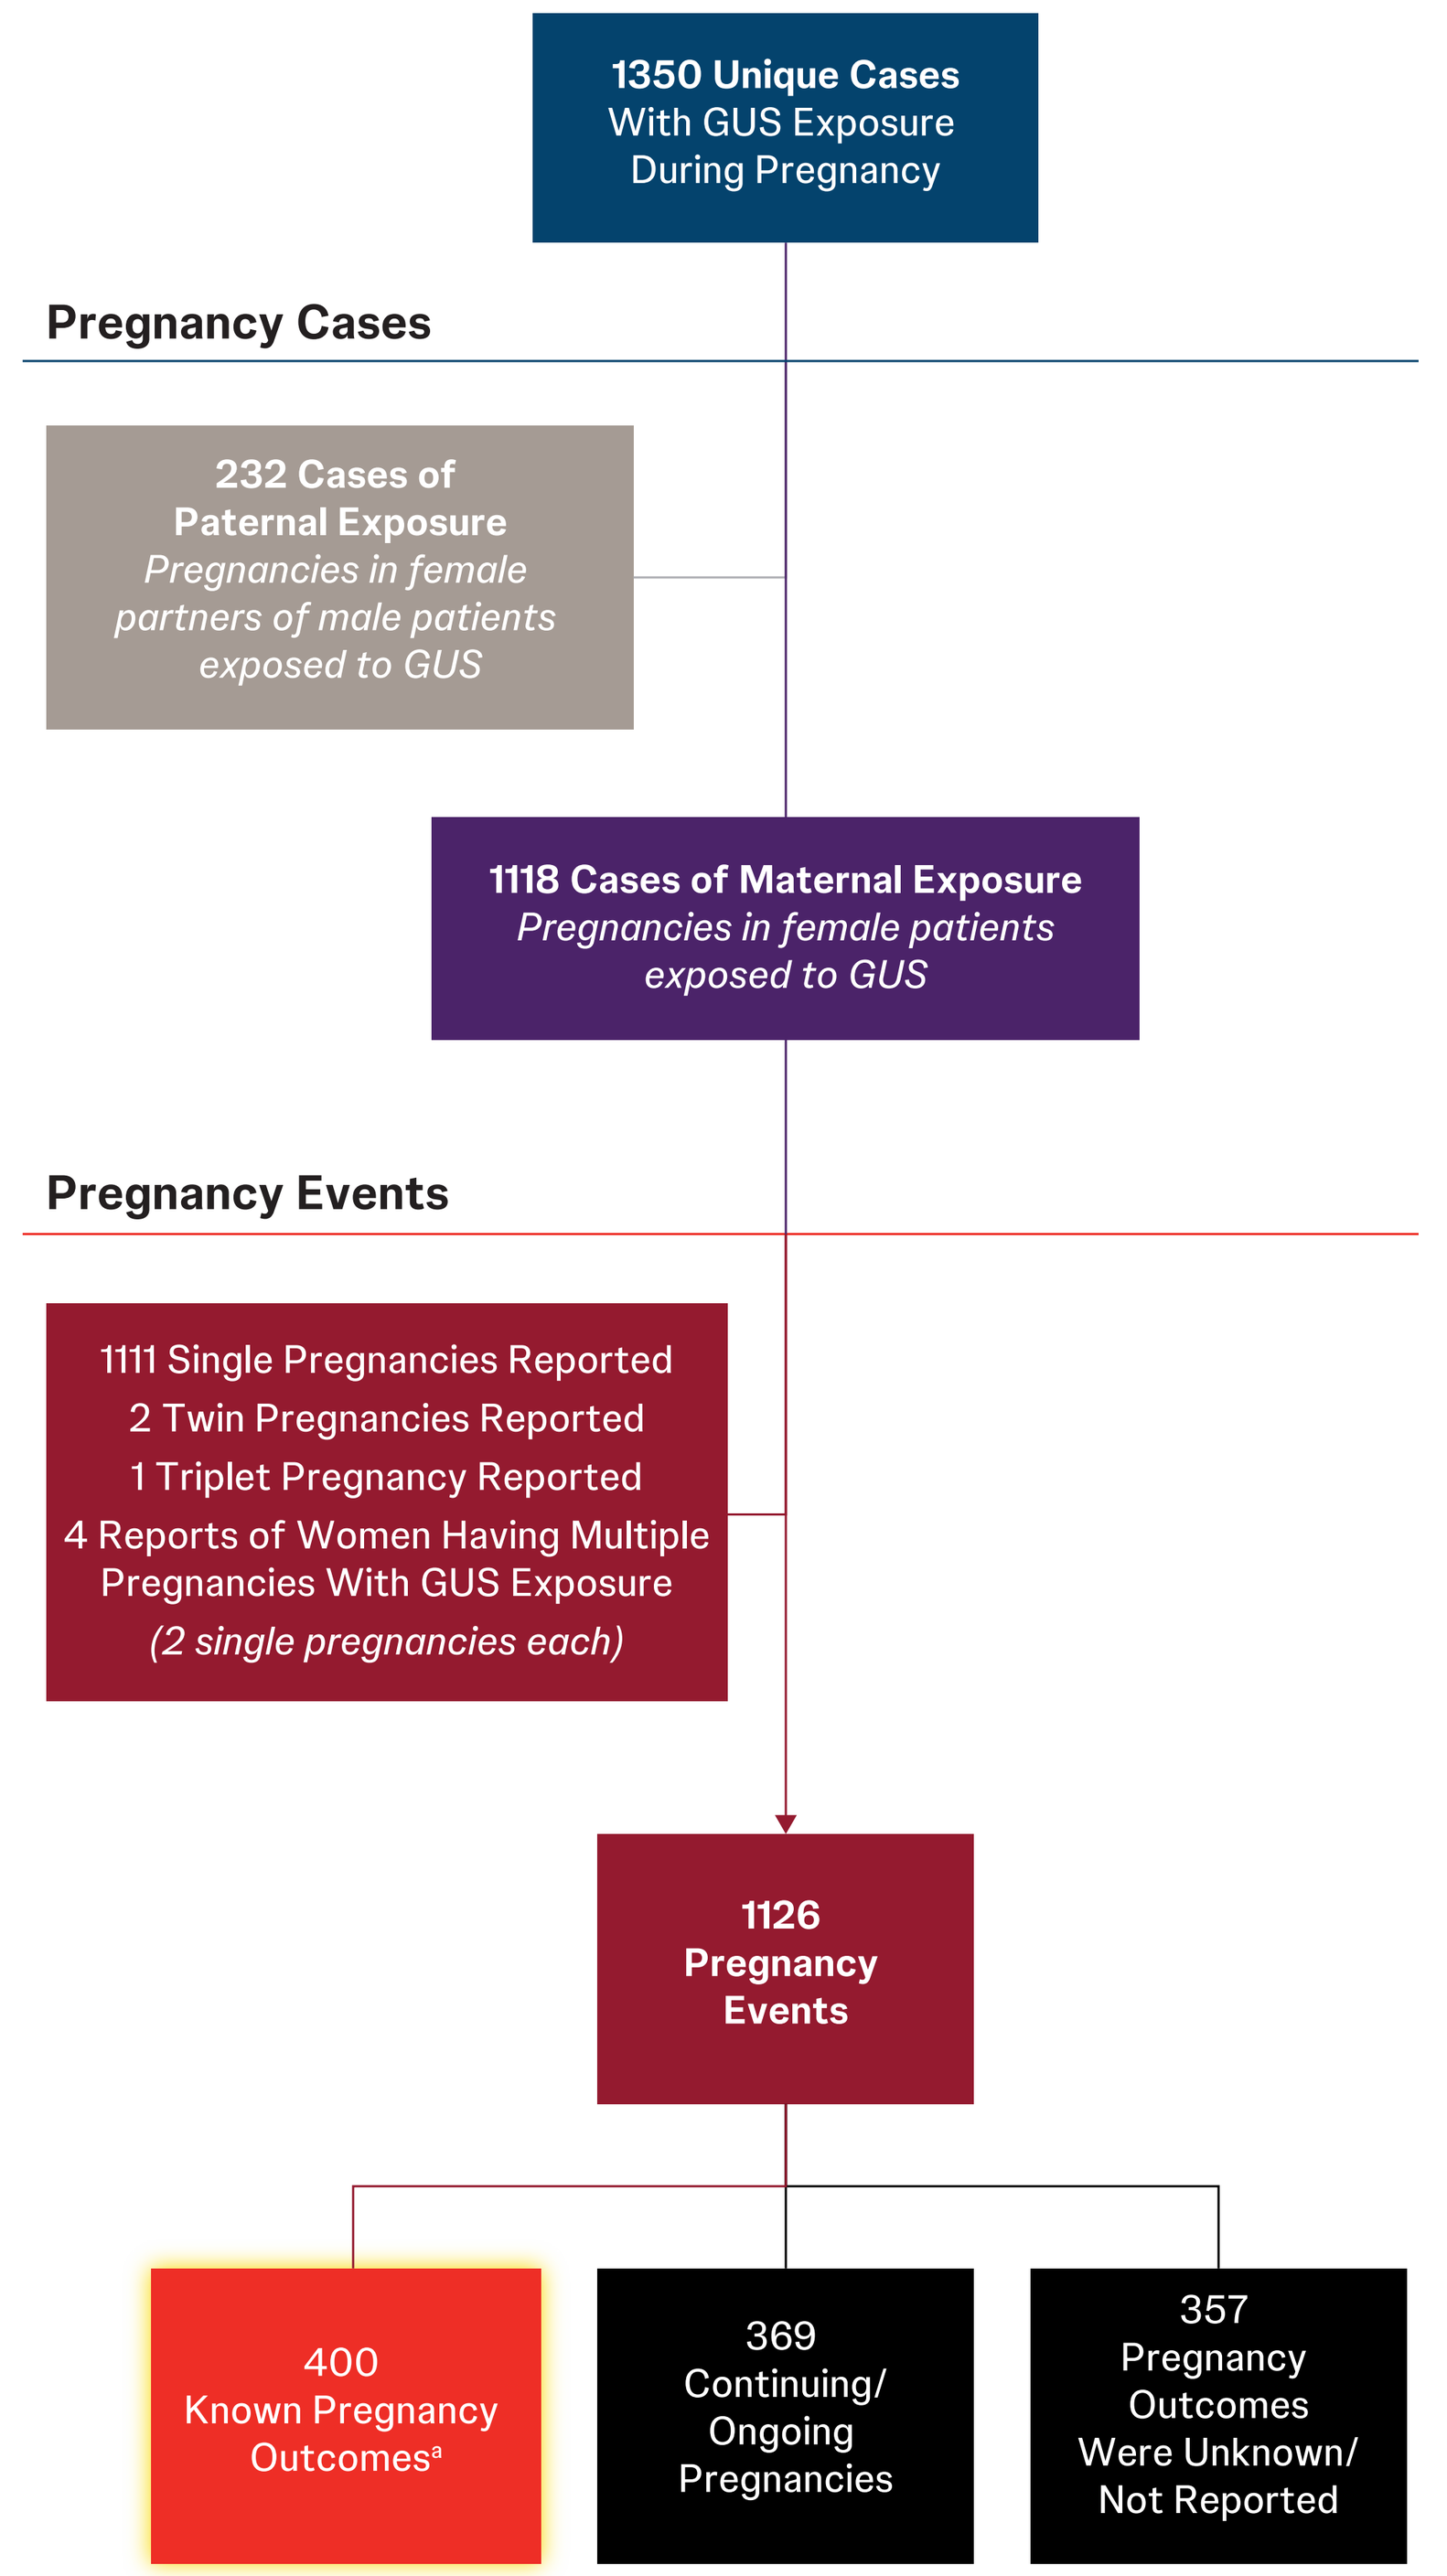
1350 Unique Cases With GUS Exposure During Pregnancy
Pregnancy Cases
232 Cases of Paternal Exposure
Pregnancies in female partners of male patients exposed to GUS
1118 Cases of Maternal Exposure
Pregnancies in female patients exposed to GUS
Pregnancy Events
1111 Single Pregnancies Reported
2 Twin Pregnancies Reported
1 Triplet Pregnancy Reported
4 Reports of Women Having Multiple Pregnancies With GUS Exposure
(2 single pregnancies each)
1126 Pregnancy Events
400 Known Pregnancy Outcomes[a]
369 Continuing/ Ongoing Pregnancies
357 Pregnancy Outcomes Were Unknown/ Not Reported

[a]Includes 287 medically confirmed pregnancies and 109 medically unconfirmed pregnancies. **GUS**=guselkumab.

Proportions of prospectively and retrospectively reported pregnancy events, N=400

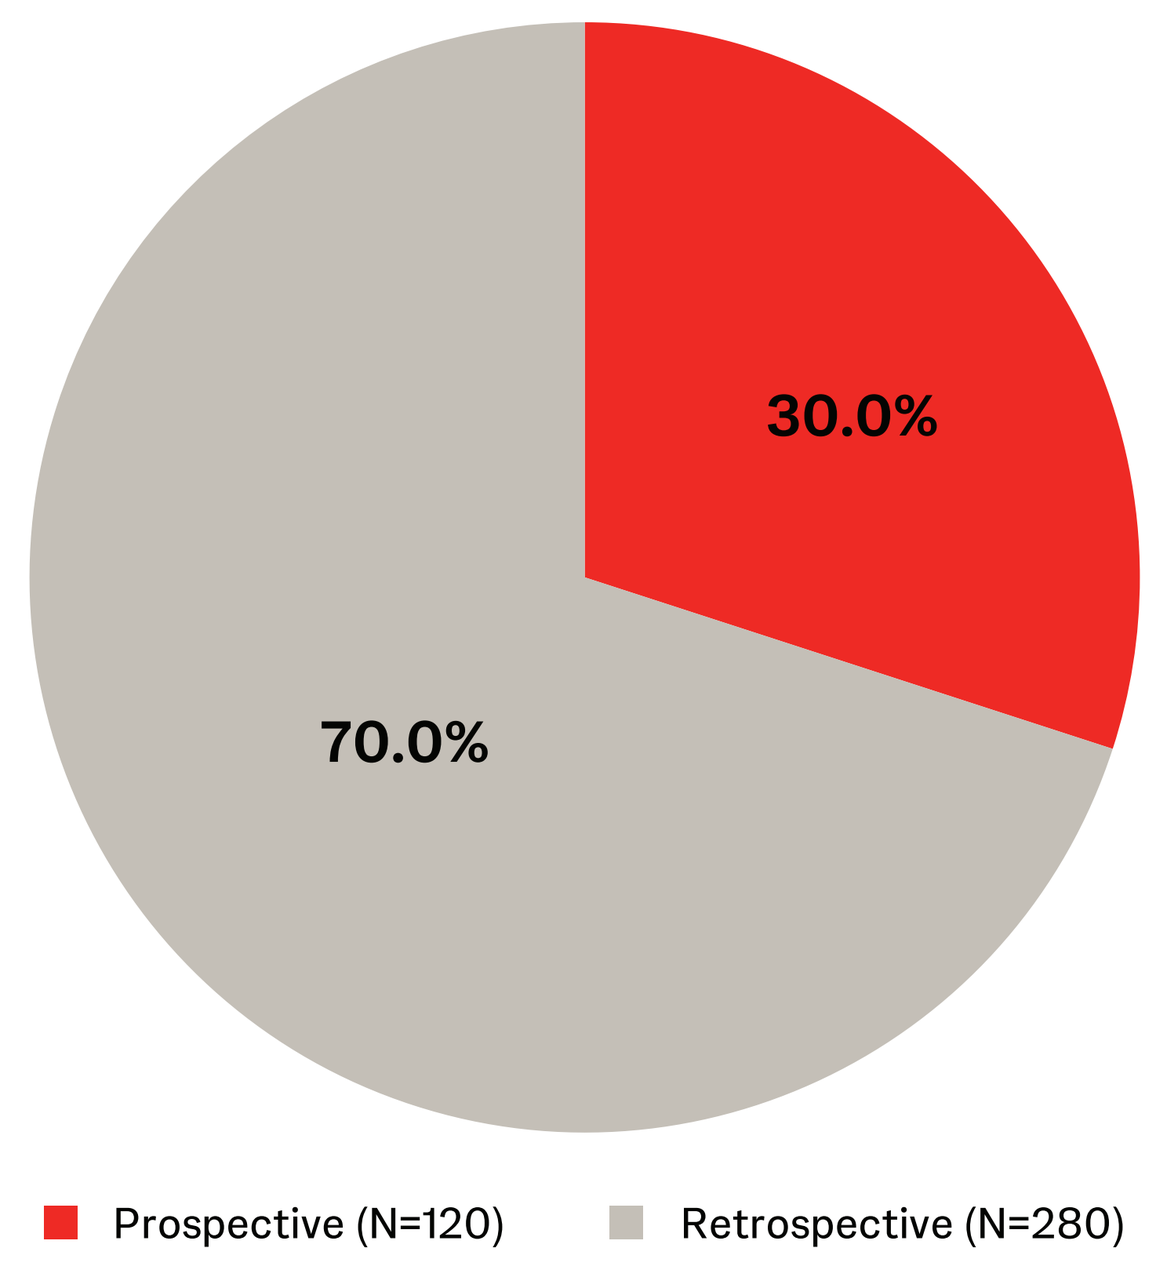
30.0%
70.0%
Prospective (N=120)
Retrospective (N=280)

Proportions of pregnancy events by reporting source, N=400

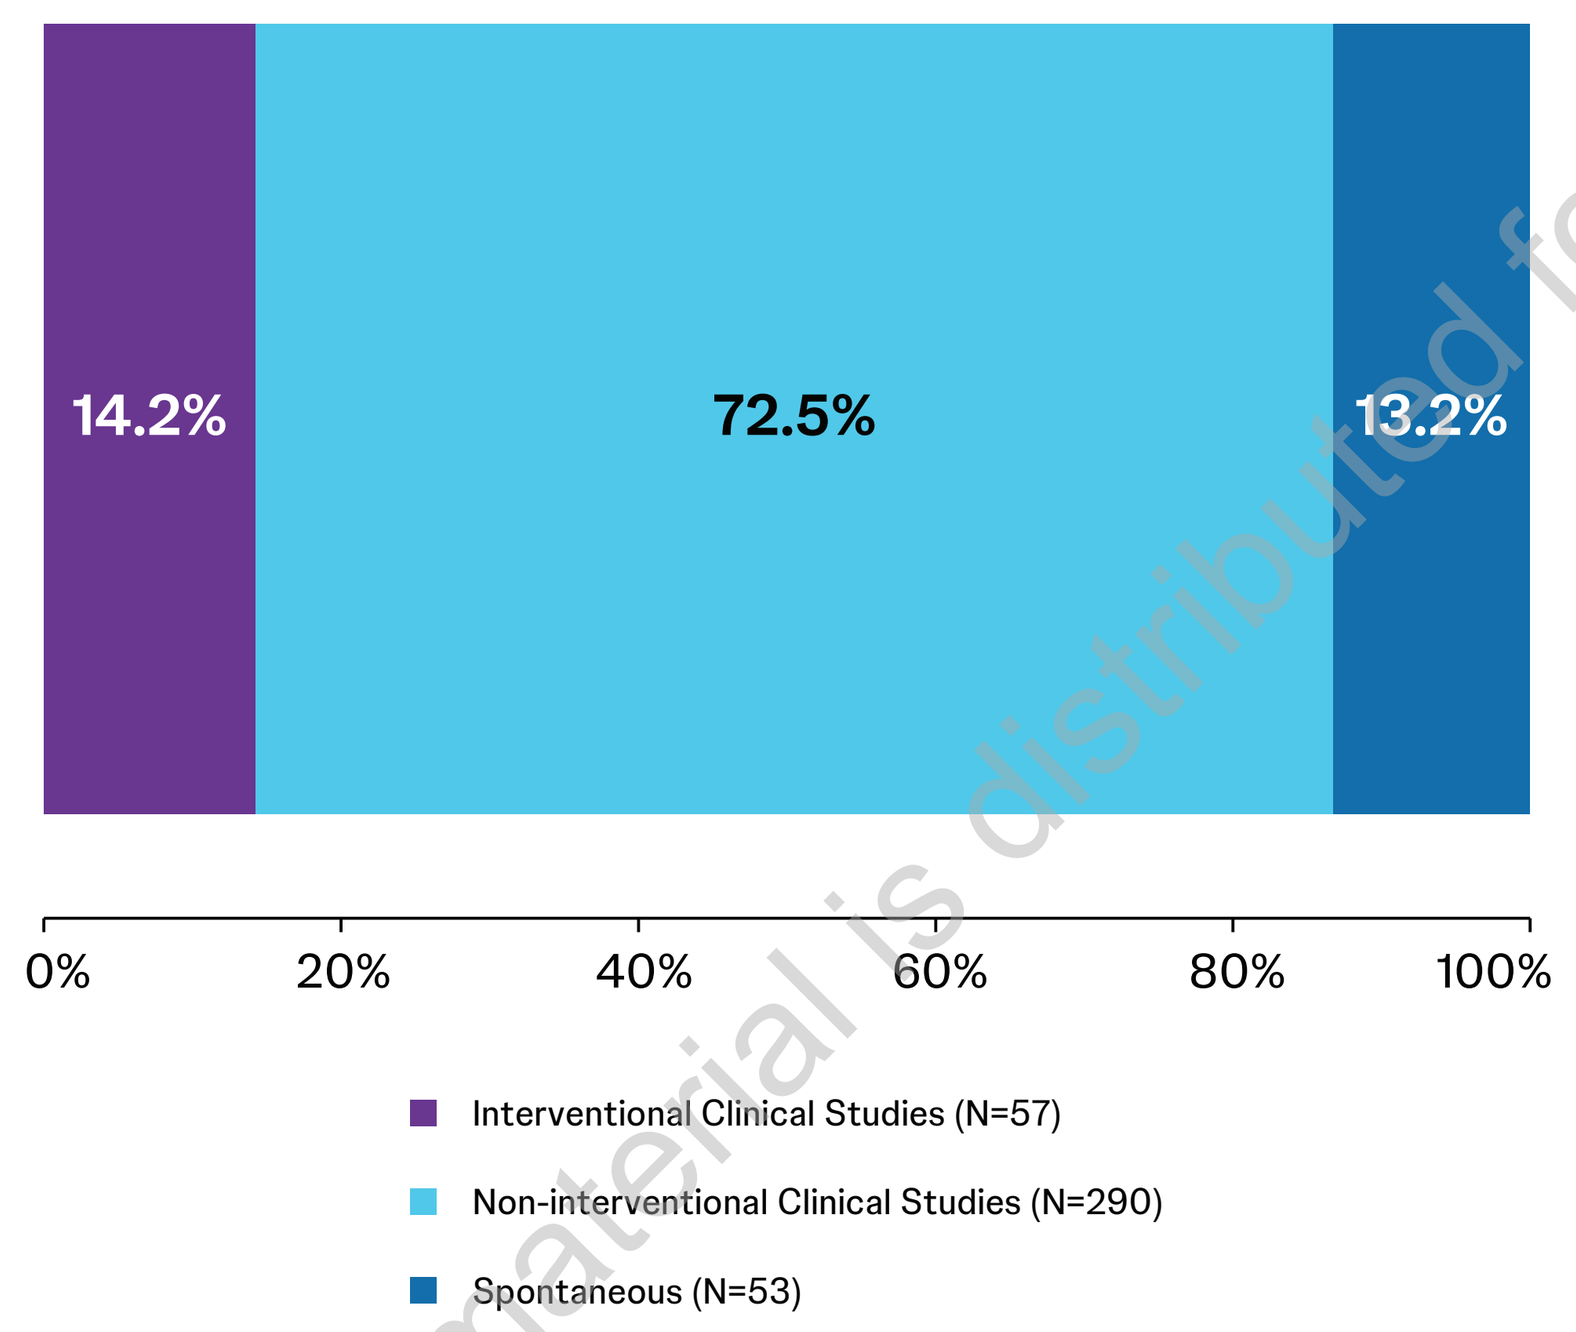
14.2%
72.5%
13.2%
0%
20%
40%
60%
80%
100%
Interventional Clinical Studies (N=57)
Non-interventional Clinical Studies (N=290)
Spontaneous (N=53)

Proportions of pregnancy cases reported by geographic region, N=396

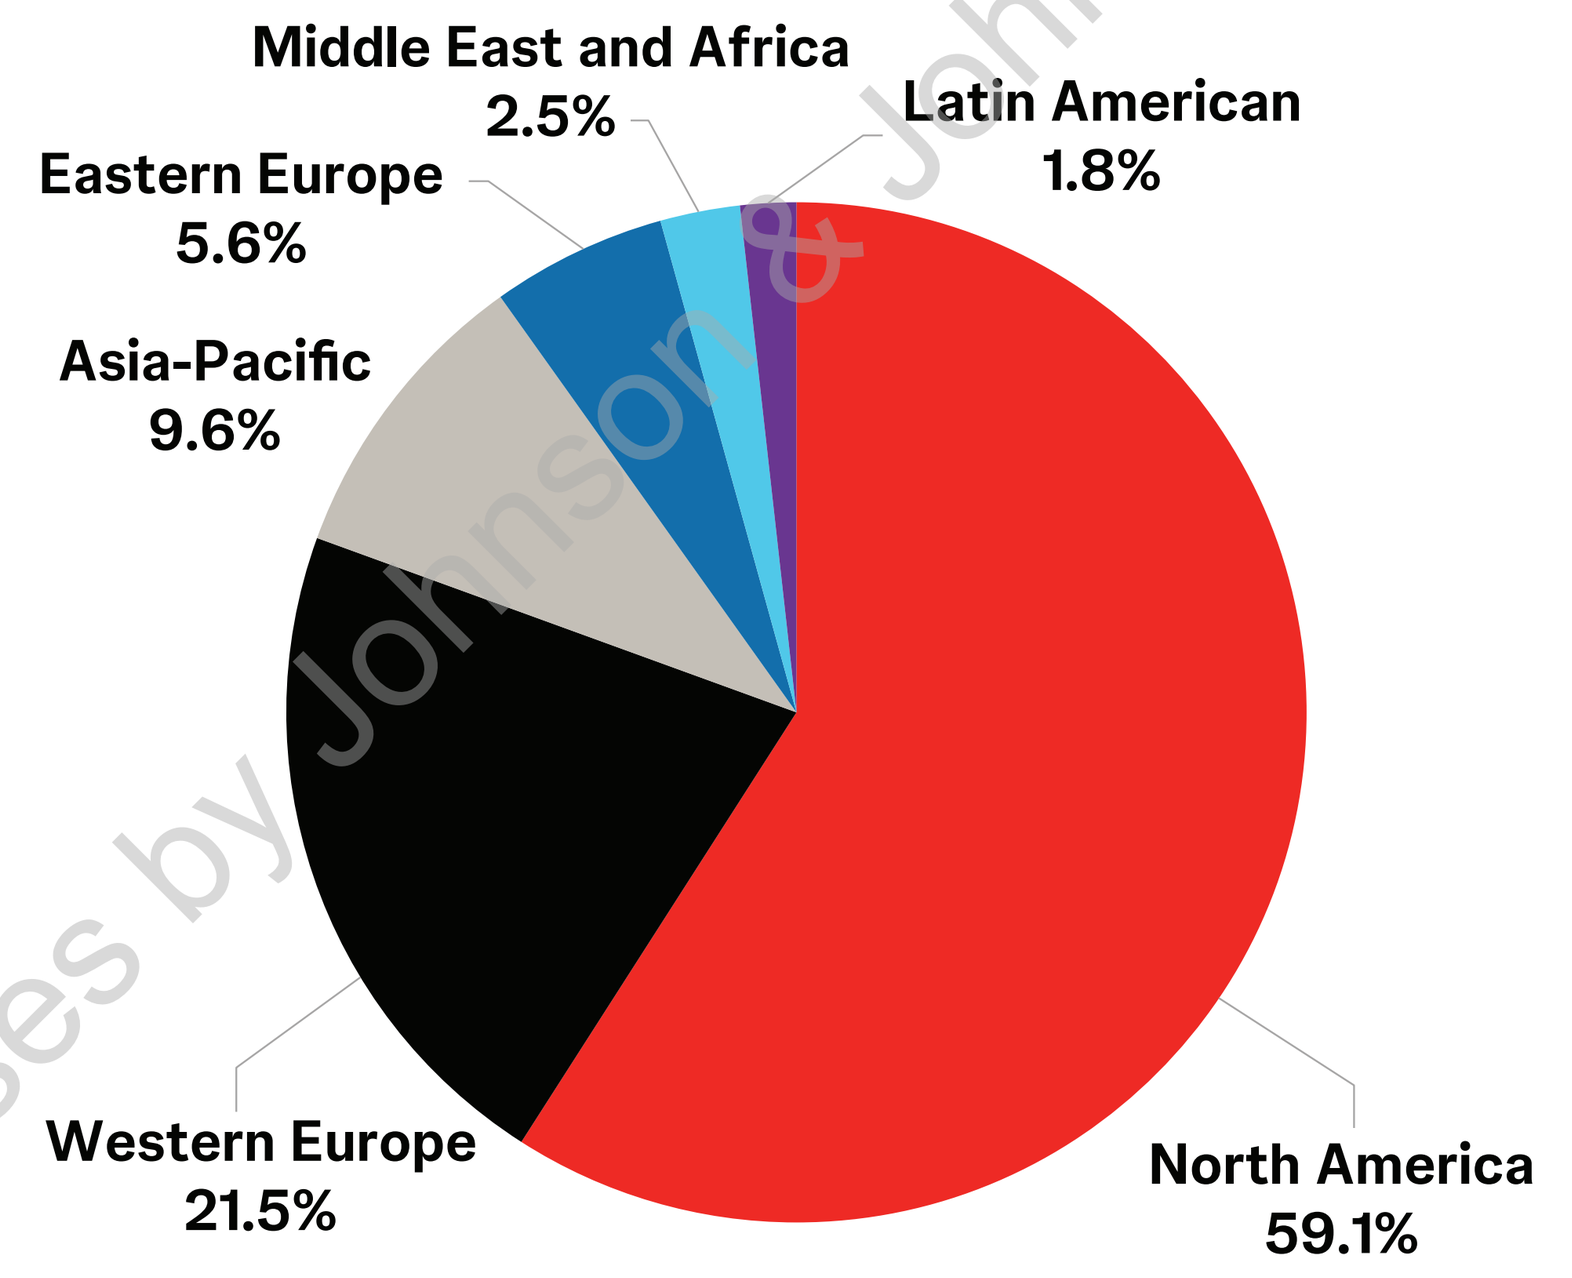
Middle East and Africa 2.5%
Latin American 1.8%
Eastern Europe 5.6%
Asia-Pacific 9.6%
Western Europe 21.5%
North America 59.1%

Proportions of pregnancy cases reported by timing of exposure, N=400

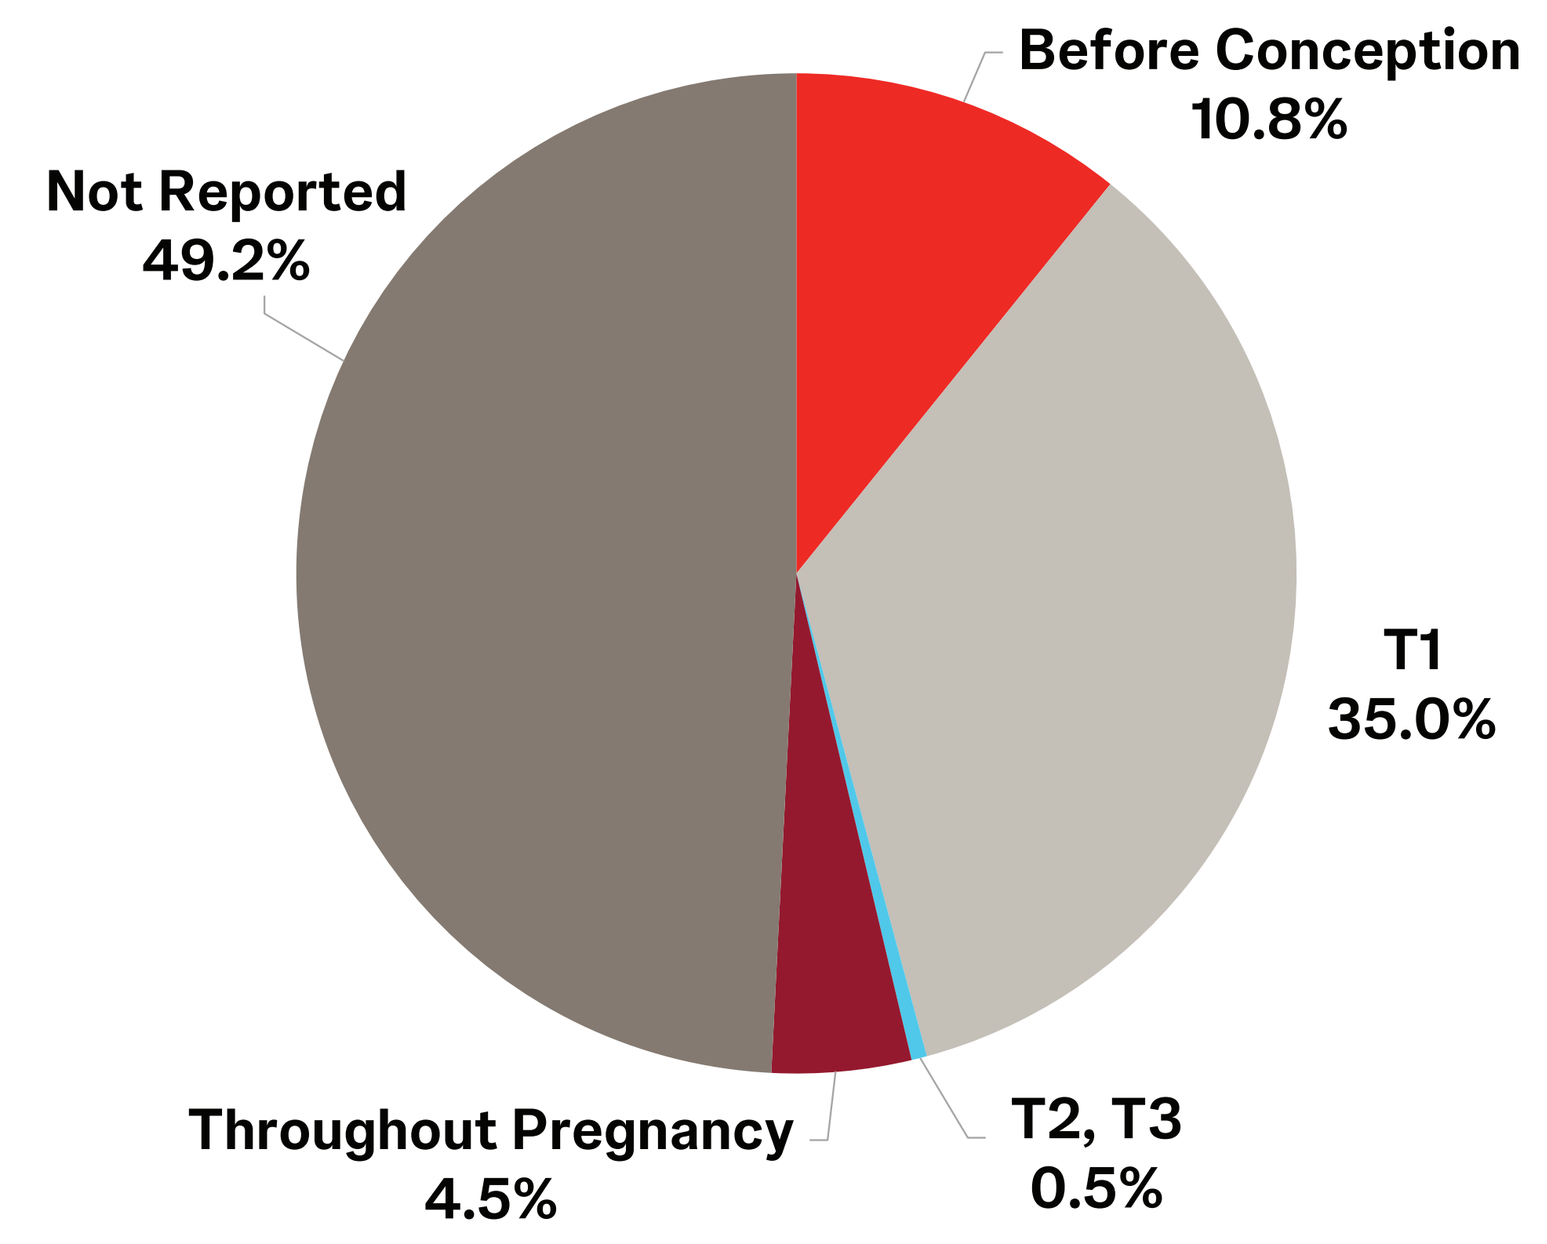
Before Conception 10.8%
Not Reported 49.2%
T1 35.0%
Throughout Pregnancy 4.5%
T2, T3 0.5%

T1=first trimester, *T2*=second trimester, *T3*=third trimester.

Proportions of pregnancy events by outcome, N=400

- 66.5% of pregnancy events with maternal exposure to GUS resulted in live birth

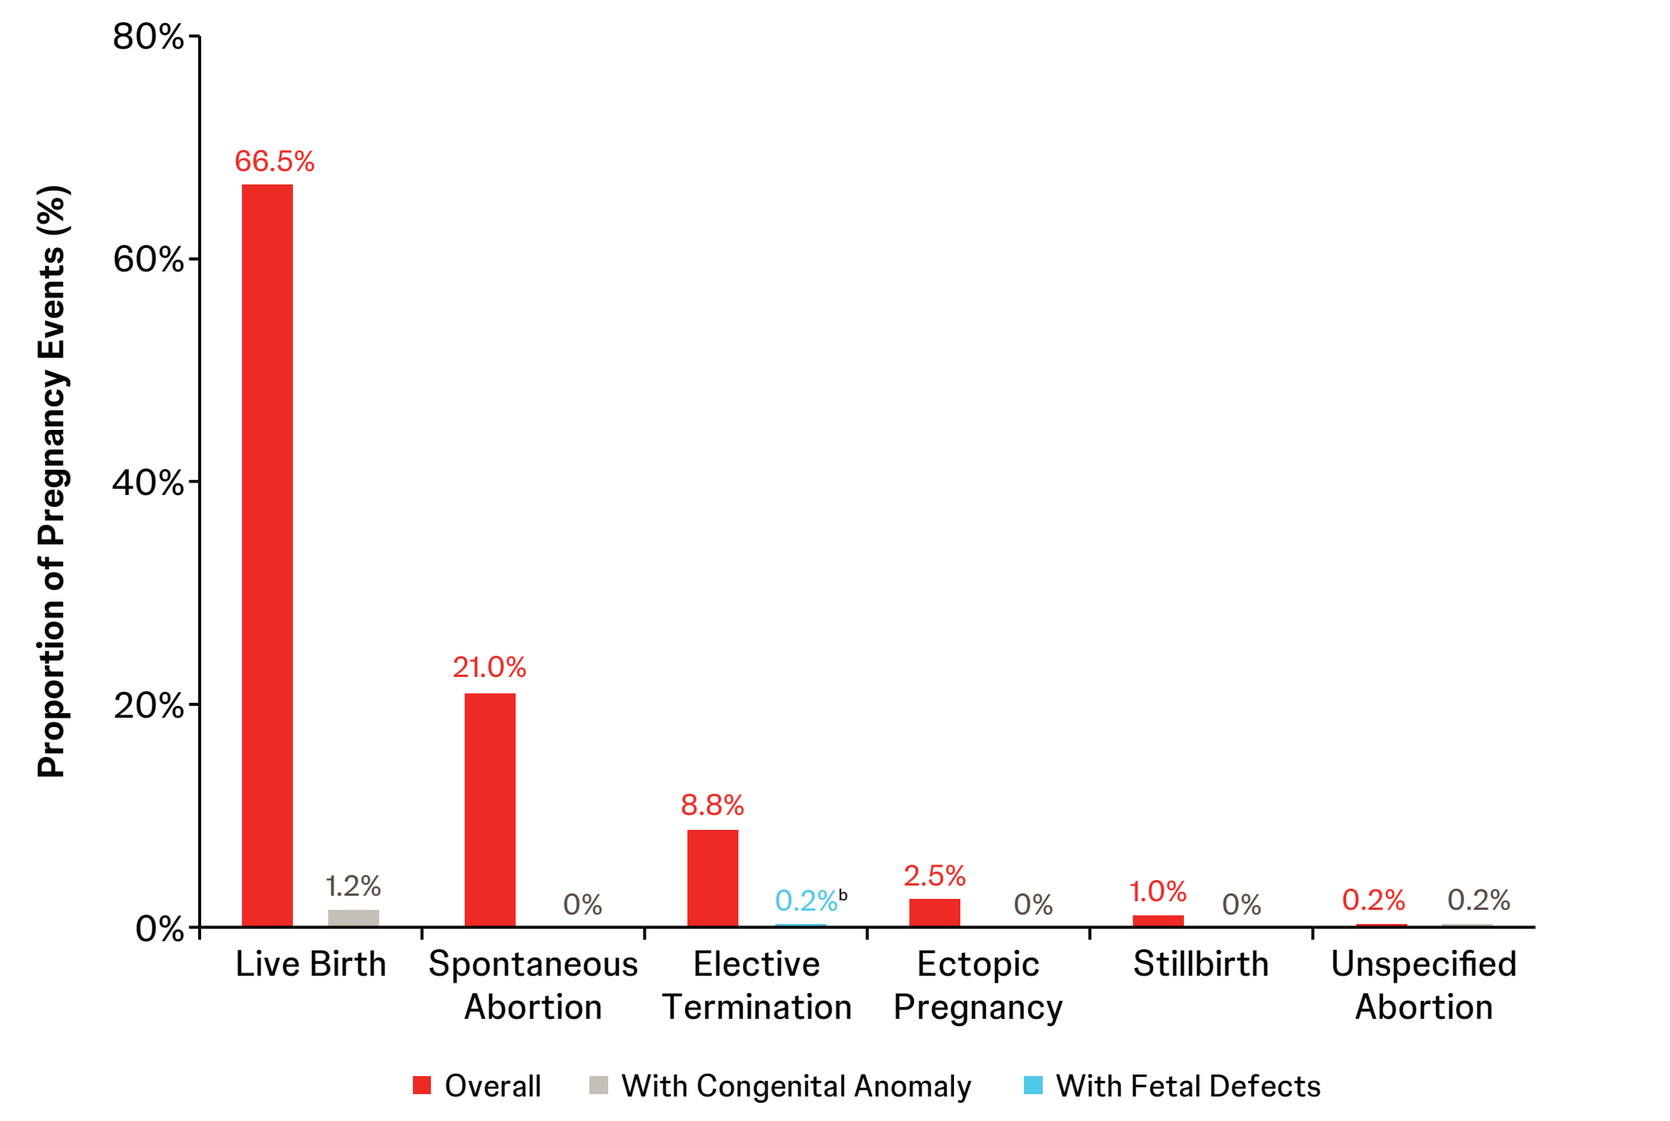
Proportion of Pregnancy Events (%)
80%
60%
40%
20%
0%
66.5%
1.2%
21.0%
0%
8.8%
0.2%[b]
2.5%
0%
1.0%
0%
0.2%
0.2%
Live Birth
Spontaneous Abortion
Elective Termination
Ectopic Pregnancy
Stillbirth
Unspecified Abortion
Overall
With Congenital Anomaly
With Fetal Defects

[b]One case reported baby adverse event of fetal disorder with no further information; conservatively, it was categorized as a fetal defect.

Rates of pregnancy outcomes with maternal exposure vs United States general population[4,5]

- Rates of pregnancy outcomes in women exposed to GUS were consistent with those of the general US population

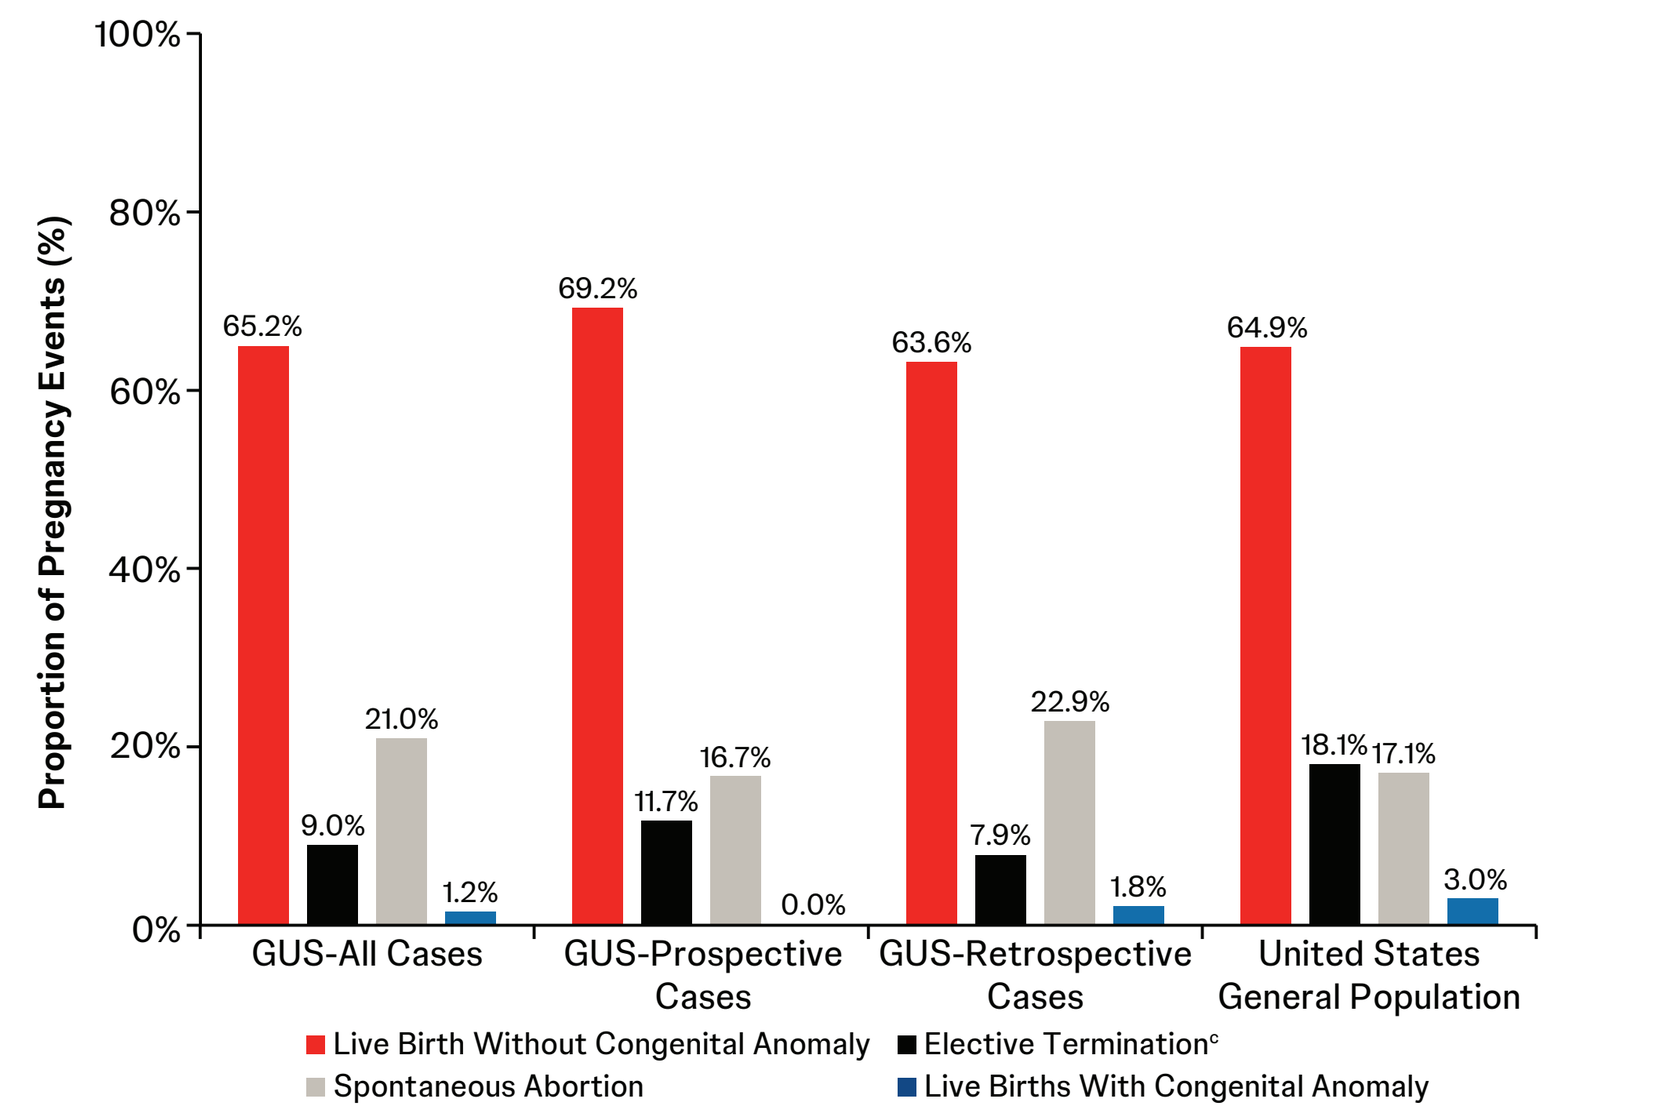
Proportion of Pregnancy Events (%)
100%
80%
60%
40%
20%
0%
65.2%
9.0%
21.0%
1.2%
69.2%
11.7%
16.7%
0.0%
63.6%
7.9%
22.9%
1.8%
64.9%
18.1%
17.1%
3.0%
GUS-All Cases
GUS-Prospective Cases
GUS-Retrospective Cases
United States General Population
Live Birth Without Congenital Anomaly
Elective Termination[c]
Spontaneous Abortion
Live Births With Congenital Anomaly

[c]Included cases reporting unspecified abortions. **GUS**=guselkumab.

Women exposed to GUS during pregnancy were treated for the following:

	Indication (N=396) n-value	Indication (N=396) Percentage
Psoriatic Disease[d]	**289**	**73.0%**
Not Reported	**70**	**17.7%**
Inflammatory Bowel Disease	**25**	**6.3%**
CD	15	3.8%
UC	10	2.5%
Other[e]	**12**	**3.0%**

[d]Includes cases reported as psoriasis, psoriatic arthritis, and psoriasis + psoriatic arthritis. [e]Other indications included hidradenitis suppurativa, palmoplantar pustulosis, guttate psoriasis, pityriasis rubra pilaris, rheumatoid arthritis, and healthy individuals from Phase 1 studies. **CD**=Crohn's disease, **GUS**=guselkumab, **UC**=ulcerative colitis.

Congenital anomalies by pregnancy outcome and timing of exposure

- Of the 400 pregnancy events with known outcomes, 6 (1.5%) pregnancies were associated with congenital anomalies

Events of Interest[f]	Number of Events	Pregnancy Outcomes	Timing of Maternal Exposure	Classification[g]
Trisomy 13[h]	1	Live Birth	Throughout pregnancy	Major anomaly Chromosomal
Congenital Heart Disease	1	Live Birth	Not reported	Major anomaly Nonchromosomal
Esophageal Atresia[i]	1	Live Birth	Not reported	Major anomaly Nonchromosomal
Cerebral Ventricle Dilation	1	Live Birth	During the first trimester	Minor anomaly Nonchromosomal
Single Umbilical Artery	1	Live Birth	During the first trimester	Minor anomaly Nonchromosomal
Fetal Malformation	1	Unspecified Abortion	During the first trimester	Termination of pregnancy due to fetal anomaly

[f]Medical Dictionary for Regulatory Activities (MedDRA, version 28.0) was used to identify adverse events based on the System Organ Class of congenital, familial, or genetic disorders, which is sub-search of the Standardized MedDRA Query of pregnancy and neonatal topics. [g]Major and chromosomal congenital anomalies per EUROCAT classification are reported unless otherwise specified. [h]Pre-term delivery at less than 37 weeks; baby died due to Trisomy 13. [i]Baby adverse event of tracheomalacia was reported. **EUROCAT**=European Surveillance of Congenital Anomalies, **MedDRA**=Medical Dictionary for Regulatory Activities.

PRESENTED BY: Allison Hester at the Congress of Clinical Rheumatology-West (CCR-West), September 17–20, 2026; Huntington Beach, California, USA. **REFERENCES: 1.** Mahadevan U, et al. *J Crohns Colitis.* 2025;19:jjaf129. **2.** Sachen KL, et al. *Front Immunol.* 2025;16:1532852. **3.** TREMFYA. Package insert. Janssen Biotech, Inc.; 2025. **4.** Curtin SC, et al. Pregnancy Rates for U.S. Women Continue to Drop. Centers for Disease Control and Prevention, U.S. Dept of Health and Human Services; 2013:1–7. **5.** About Birth Defects. U.S. Centers for Disease Control and Prevention. February 25, 2025. **ACKNOWLEDGMENTS:** Medical writing support was provided by Alyssa Cortina, PharmD, under the direction of the authors in accordance with Good Publication Practice guidelines (*Ann Intern Med.* 2022;175:1298–1304). Layout design and formatting for this encore presentation were provided by Amit Kavle (SIRO Medical Writing Pvt. Ltd., India). This study and presentation were sponsored by Johnson & Johnson. **DISCLOSURES: UM** has served as a consultant and an advisory board member for AbbVie, Allergan, Bristol Myers Squibb, Gilead, Johnson & Johnson, and Takeda; has received research funding from Celgene, Genentech, and Pfizer. **ML** has reported research support from Celltrion, Eli Lilly, Pfizer, and Takeda; and has consulted for AbbVie, Bristol Myers Squibb, Celltrion, Intercept, Johnson & Johnson, Merck, Lilly, Pfizer, Prometheus, Roivant, Sanofi, Spyre, Target RWE, and Takeda. **MJ** has reported research grants for investigator-driven studies from Novo Nordisk Foundation and Takeda (grant no. NNF23OC0081717); has consulted for Ferring, Orion Pharma, and Takeda; has received speaker's fees from Eli Lilly, Ferring, MSD, Takeda, and Tillotts Pharma; and is on the advisory board of AbbVie, Eli Lilly, PharmaCosmos, and Tillotts Pharma. **CL, AG, MRB, HL,** and **AH** are employees of Johnson & Johnson and may own stock/stock options in Johnson & Johnson. **JPG** has received grants from AbbVie, Biogen, Casen Fleet, Celgene/Bristol Myers Squibb, Chiesi, Dr. Falk Pharma, Faes Farma, Ferring, Gebro Pharma, Gilead/Galapagos/Alfasigma, Johnson & Johnson, Kern Pharma, Eli Lilly, MSD, Mylan, Norgine, Italfarmaco, Otsuka Pharmaceutical, Pfizer, Roche, Sandoz, Takeda, Sanofi, Shire Pharmaceuticals, STADA, Teva, Tillotts Pharma, and Vifor Pharma. **MC** has received grants from AbbVie, Biogen, Johnson & Johnson, Eli Lilly, and Pfizer; has received fees from AbbVie, Faes, Johnson & Johnson, and Pfizer. **PREVIOUSLY PRESENTED AT:** The European Crohn's and Colitis Organisation (ECCO); February 18–21, 2026; Stockholm, Sweden.