Patient-Reported Outcomes Through 1 Year With Guselkumab in Participants With Active PsA and Inadequate Response to 1 Prior TNFi in SOLSTICE

Alexis Ogdie[1], Alice B. Gottlieb[2], Philip J. Mease[3,4], Joseph F. Merola[5], Elizabeth Adamson[6], Soumya D. Chakravarty[6,7], Yanli Wang[6], Fowzia Ibrahim[8], Kimberly Parnell Lafferty[6], Christopher T. Ritchlin[9], Jose U. Scher[10]

[1]Division of Rheumatology, University of Pennsylvania School of Medicine, Philadelphia, PA, USA; [2]Department of Dermatology, UT Southwestern Medical Center, Dallas, TX, USA; [3]Rheumatology Research, Providence Swedish Medical Center, Seattle, WA, USA; [4]University of Washington School of Medicine, Seattle, WA, USA; [5]Department of Dermatology and Department of Medicine, Division of Rheumatology, UT Southwestern Medical Center and O'Donnell School of Public Health, Dallas, TX, USA; [6]Johnson & Johnson, Horsham, PA, USA; [7]Division of Rheumatology, Drexel University College of Medicine, Philadelphia, PA, USA; [8]Johnson & Johnson, High Wycombe, UK; [9]Department of Medicine, Allergy/Immunology, and Rheumatology, University of Rochester Medical Center, Rochester, NY, USA; [10]New York University Grossman School of Medicine, New York, NY, USA

Scan the QR code.
The QR code is intended to provide scientific information for individual reference, and the information should not be altered or reproduced in any way.

Key Takeaways

- **At W24 in SOLSTICE, GUS-treated participants with active PsA had greater improvements vs PBO in overall HRQoL as assessed by PROMIS-29 and PsAID-12**
- **Participants receiving GUS also had greater response rates vs PBO at W24 for achieving PsAID-12 meaningful improvement and PASS**
- **Mean improvements and response rates were maintained through W52 among GUS-randomized participants**

Background

- Psoriatic arthritis (PsA) is a chronic inflammatory disease with significant impacts on health-related quality of life (HRQoL)[1]
- Many patients treated with tumor necrosis factor inhibitors (TNFi) experience lack or loss of efficacy[1,2]
 - There are no clear guidelines for subsequent therapy options[1,2]
- Guselkumab (GUS) is a fully human, dual-acting, selective interleukin (IL)-23p19-subunit inhibitor[3]
 - GUS demonstrated efficacy in participants with active PsA and inadequate response (inadequate efficacy and/or intolerance) to 1 prior TNFi (TNFi-IR) through Week (W) 24 in the phase 3b double-blind, randomized, placebo (PBO)-controlled SOLSTICE study[4]

Objective

To evaluate the effect of GUS on patient-reported outcomes (PROs) in participants with TNFi-IR in SOLSTICE

Methods

SOLSTICE – Study Design

Key Inclusion Criteria

- Age ≥18 years
- Active PsA (≥3 SJC; ≥3 TJC; CRP ≥0.3 mg/dL); CASPAR criteria met
- **TNFi-IR to 1 prior TNFi**
- History of or active PsO (≥1 plaque ≥2 cm and/or nail PsO)

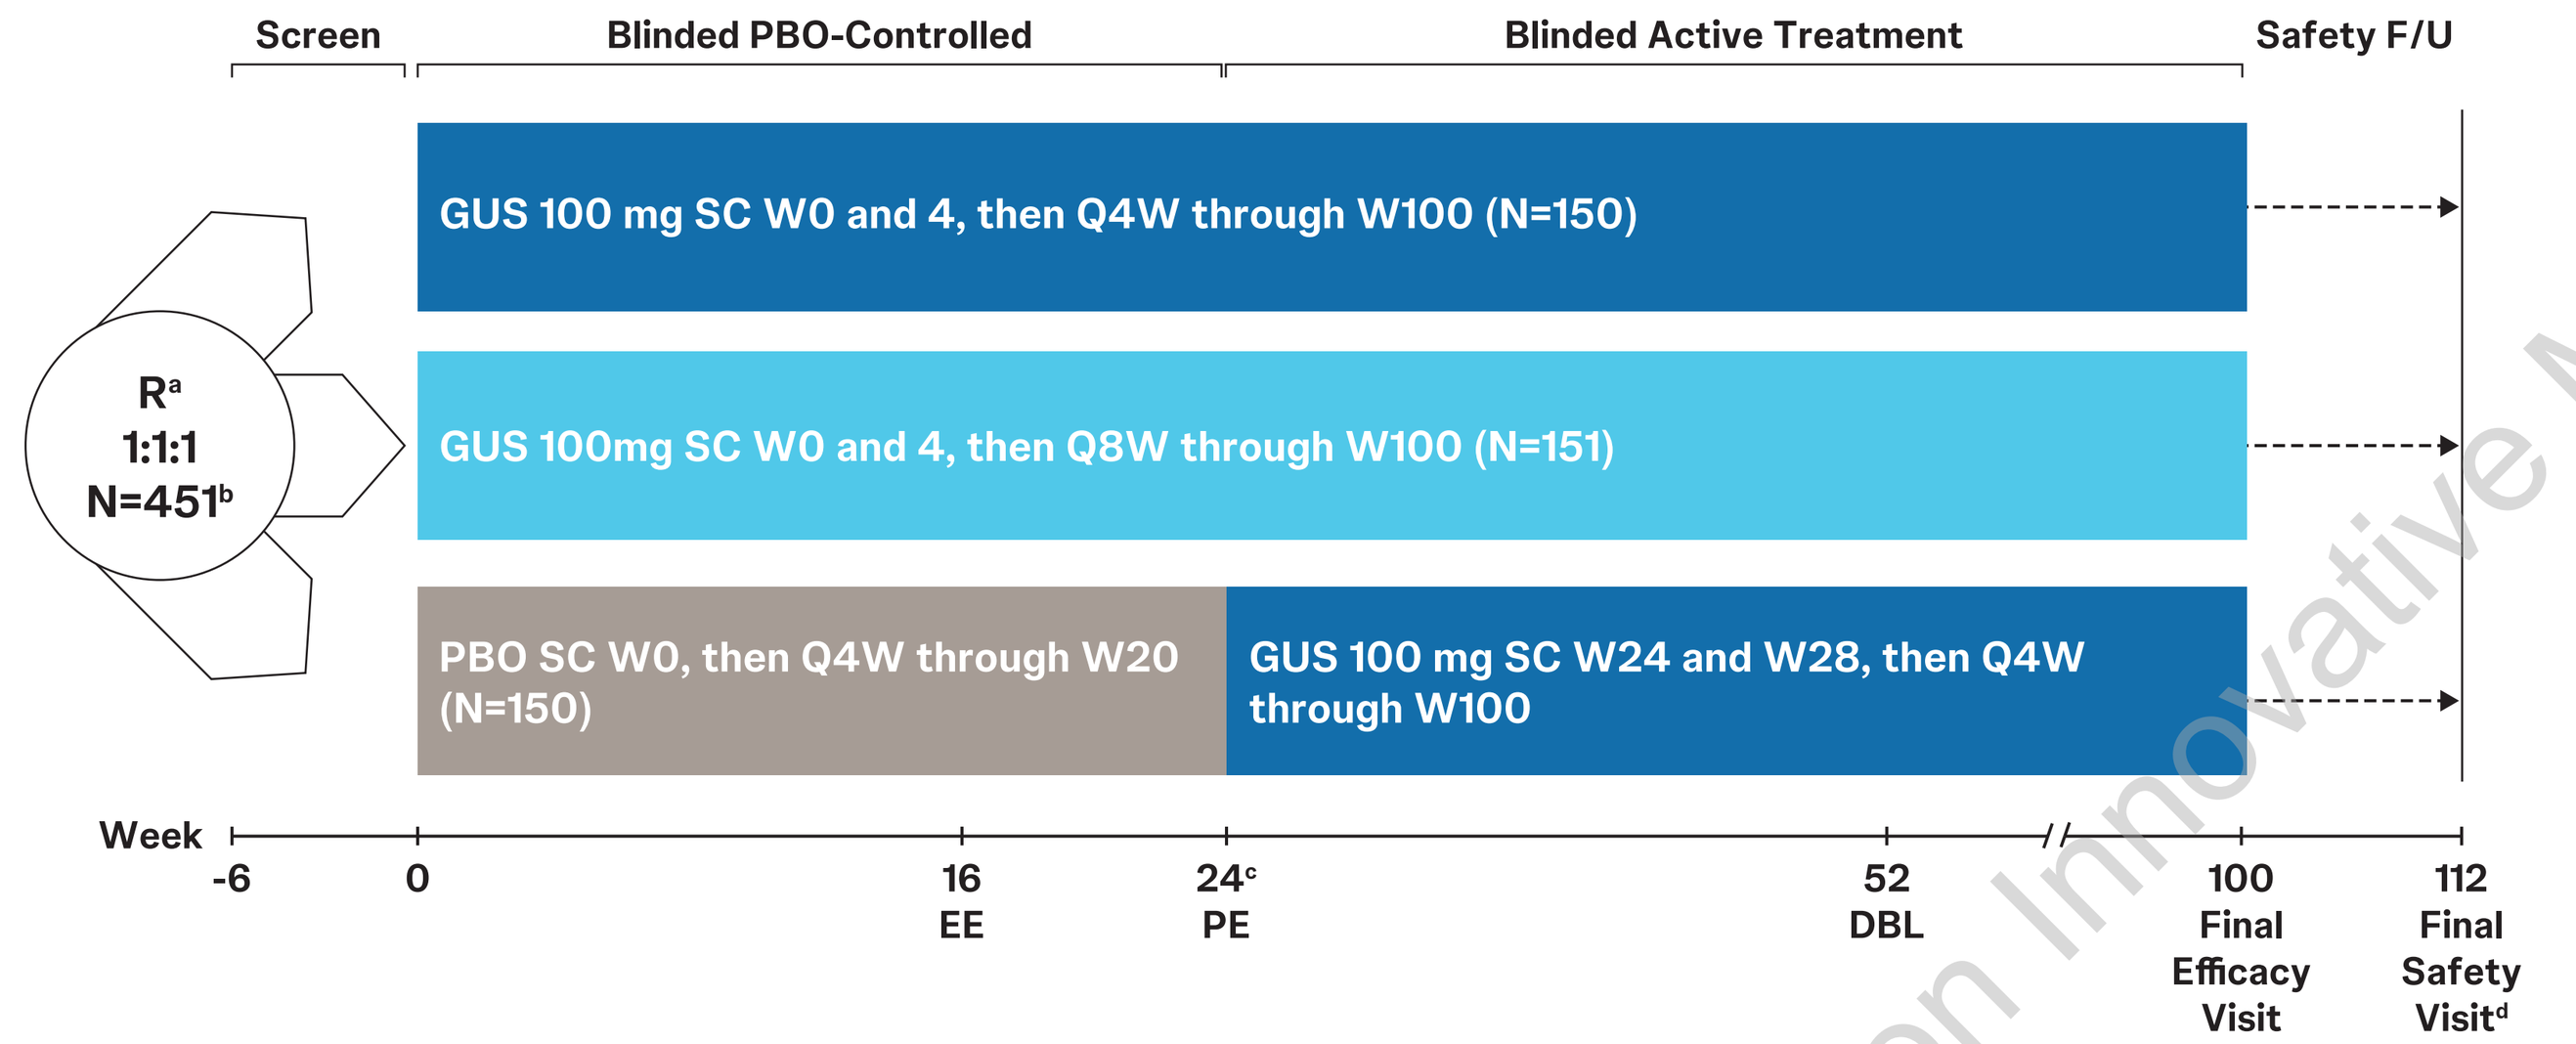

Screen
Blinded PBO-Controlled
Blinded Active Treatment
Safety F/U
R[a]
1:1:1
N=451[b]
GUS 100 mg SC W0 and 4, then Q4W through W100 (N=150)
GUS 100mg SC W0 and 4, then Q8W through W100 (N=151)
PBO SC W0, then Q4W through W20 (N=150)
GUS 100 mg SC W24 and W28, then Q4W through W100
Week
-6
0
16
EE
24[c]
PE
52
DBL
100
Final Efficacy Visit
112
Final Safety Visit[d]

PRO Assessments:

- PROMIS-29: 7 domains (physical function, social participation, depression, anxiety, pain interference, fatigue, and sleep disturbance) and a pain intensity VAS; domain scores are converted to standardized T-scores
- PsAID-12: 12 domains examining physical and psychological perspectives (final score range, 0 [no difficulty] to 10 [extreme difficulty])

Statistical Analyses:

- LSM changes from baseline in PROMIS-29 domains and total PsAID-12 score, and proportions of participants achieving PsAID-12 MCID (improvement ≥3) and PASS (score ≤4) were determined at W24 and W52

*[a]Randomization was stratified by baseline use of csDMARDs. [b]Total number randomized=453, the full analysis set of 451 excludes 1 participant who was double randomized. [c]Crossover. [d]Final safety F/U at W112 is 12W after final study agent administration. **CASPAR**=ClASsification criteria for Psoriatic ARthritis, **CRP**=C-reactive protein, **csDMARDs**=conventional synthetic disease-modifying antirheumatic drugs, **DBL**=database lock, **EE**=early escape, **F/U**=follow-up, **LSM**=least-squares mean, **MCID**=minimal clinically important difference, **PASS**=Patient Acceptable Symptom State, **PE**=primary endpoint, **PROMIS-29**=Patient-Reported Outcomes Measurement Information System-29 profile instrument, **PsAID-12**=12-item PsA Impact of Disease, **PsO**=psoriasis, **Q4/8W**=every 4/8 weeks, **SC**=subcutaneous, **SJC**=swollen joint count, **TJC**=tender joint count, **VAS**=visual analog scale.*

Results

Baseline characteristics were well balanced across treatment groups

	GUS 100 mg Q4W (N = 150)	GUS 100 mg Q8W (N = 151)	PBO (N = 150)
Demographics			
Age, yrs	50.6 (13.3)	51.9 (12.9)	49.2 ± 12.6
Female	50%	51%	57%
Race, White	94%	95%	94%
BMI, kg/m²	30.0 (6.5)	30.9 (6.6)	30.0 (7.3)
Disease Characteristics			
SJC (0-66)	10.7 (7.8)	10.3 (6.6)	10.2 (6.4)
TJC (0-68)	18.1 (12.6)	17.1 (11.2)	16.8 (11.6)
PROMIS-29			
Physical function T-score[a]	38.5 (6.1)	38.0 (5.9)	38.2 (5.4)
Social participation T-score[a]	45.3 (8.1)	45.8 (8.7)	45.0 (7.2)
Depression T-score[b]	51.7 (9.2)	51.9 (9.4)	52.8 (9.7)
Anxiety T-score[b]	53.9 (10.2)	52.5 (9.4)	54.4 (10.4)
Pain interference T-score[b]	62.1 (6.0)	61.9 (6.4)	62.6 (5.9)
Fatigue T-score[b]	55.5 (8.8)	55.2 (9.7)	55.9 (9.4)
Sleep disturbance T-score[b]	53.5 (6.9)	52.7 (7.9)	53.5 (8.3)
Pain intensity VAS (0-10)[b]	6.6 (1.9)	6.5 (1.9)	6.6 (1.8)
PsAID-12 (0-10)	5.6 (1.9)	5.5 (2.0)	5.7 (1.9)
Concomitant methotrexate use	50%	46%	48%

*Data shown are mean (SD) unless otherwise noted. [a]Higher scores indicate better health outcomes. [b]Higher scores indicate more severe symptoms. **BMI**=body mass index, **SD**=standard deviation.*

LSM improvements from baseline in PROMIS-29 T-scores and pain intensity VAS were greater for GUS-treated participants vs PBO at W24 across all domains

LSM improvement from baseline at W24 and W52 in PROMIS-29 domain T-scores and pain intensity VAS

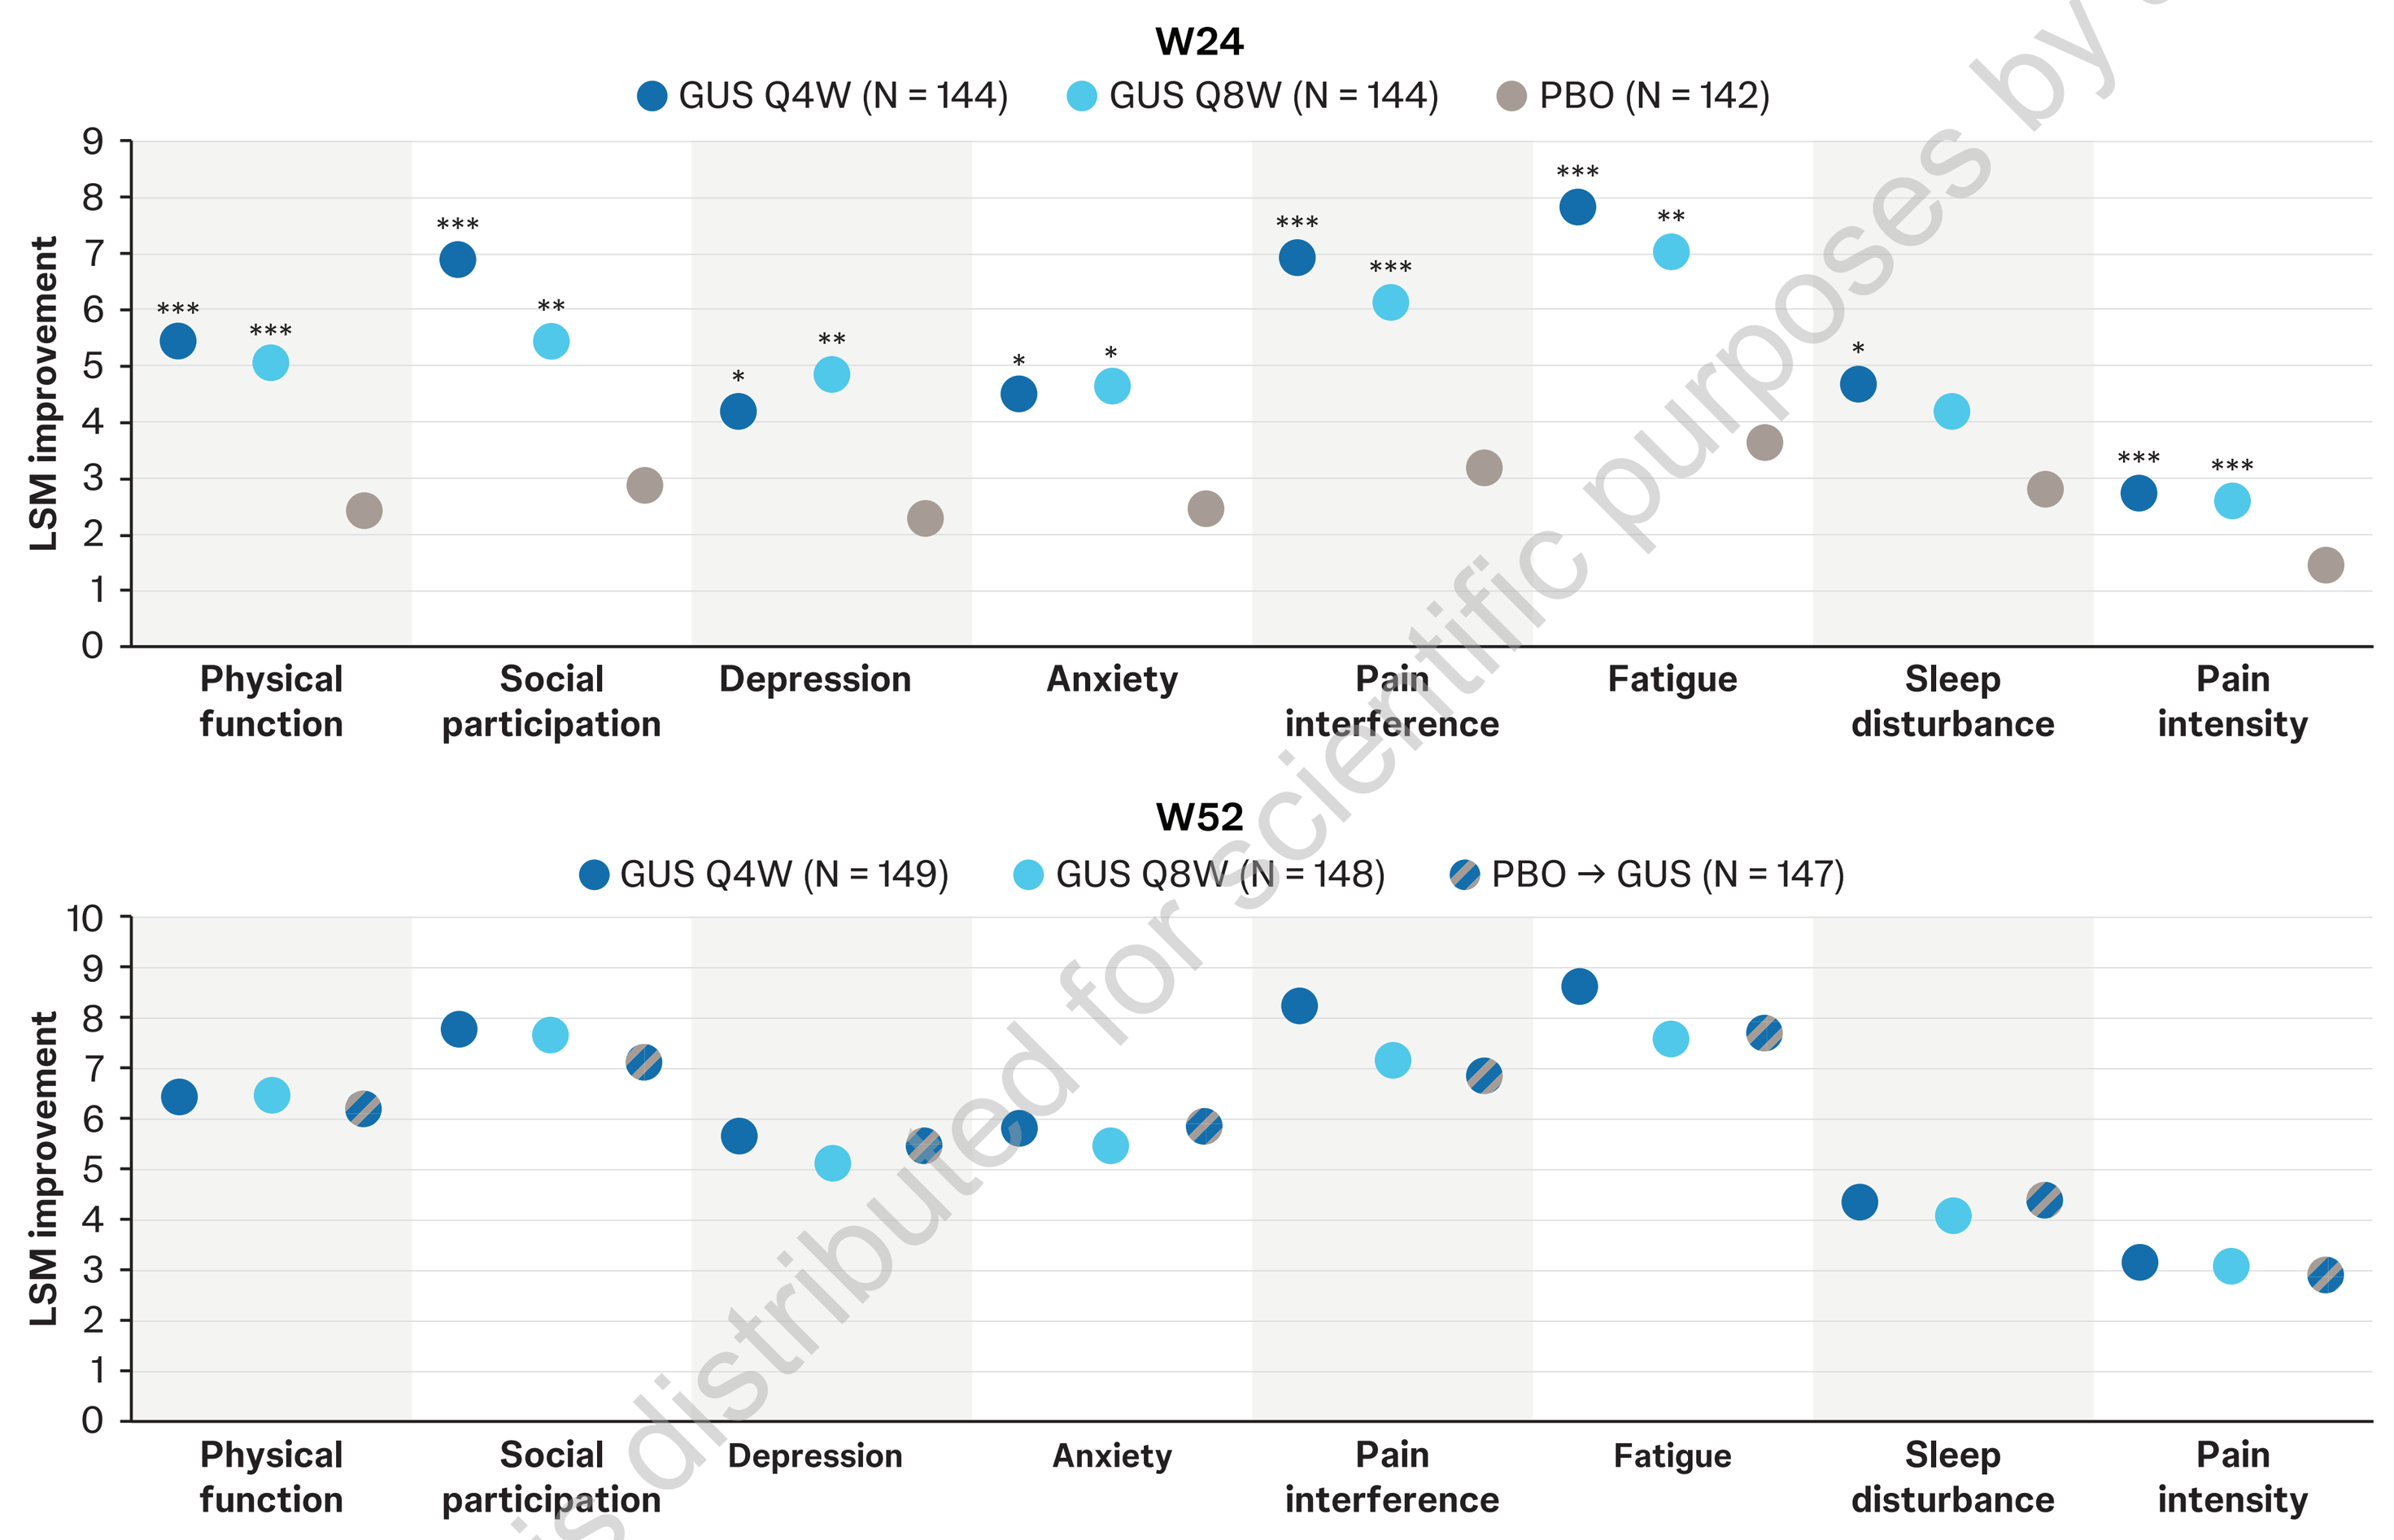

W24
GUS Q4W (N = 144)
GUS Q8W (N = 144)
PBO (N = 142)
LSM improvement
Physical function
Social participation
Depression
Anxiety
Pain interference
Fatigue
Sleep disturbance
Pain intensity
W52
GUS Q4W (N = 149)
GUS Q8W (N = 148)
PBO → GUS (N = 147)

Nominal *p<0.05, **p<0.01, *p<0.001**

LSM and nominal p values were based on Mixed-Effect Model for Repeated Measures. Participants were considered nonresponders through W24 if they increased dose/initiated csDMARDs or oral corticosteroids or initiated protocol-prohibited PsA therapies and through W52 if they discontinued study agent for any reason other than natural disaster (COVID-19 pandemic)/major disruption (Ukraine-Russia crisis). Data impacted by natural disaster/major disruption were not used; other missing data were accounted for in the analysis model.

LSM changes in PsAID-12 total scores showed greater improvement from baseline with both GUS dosing regimens vs PBO at W24

- Improvements with GUS were maintained or numerically increased through W52

LSM improvements from baseline in PsAID-12 total scores at W24 and W52

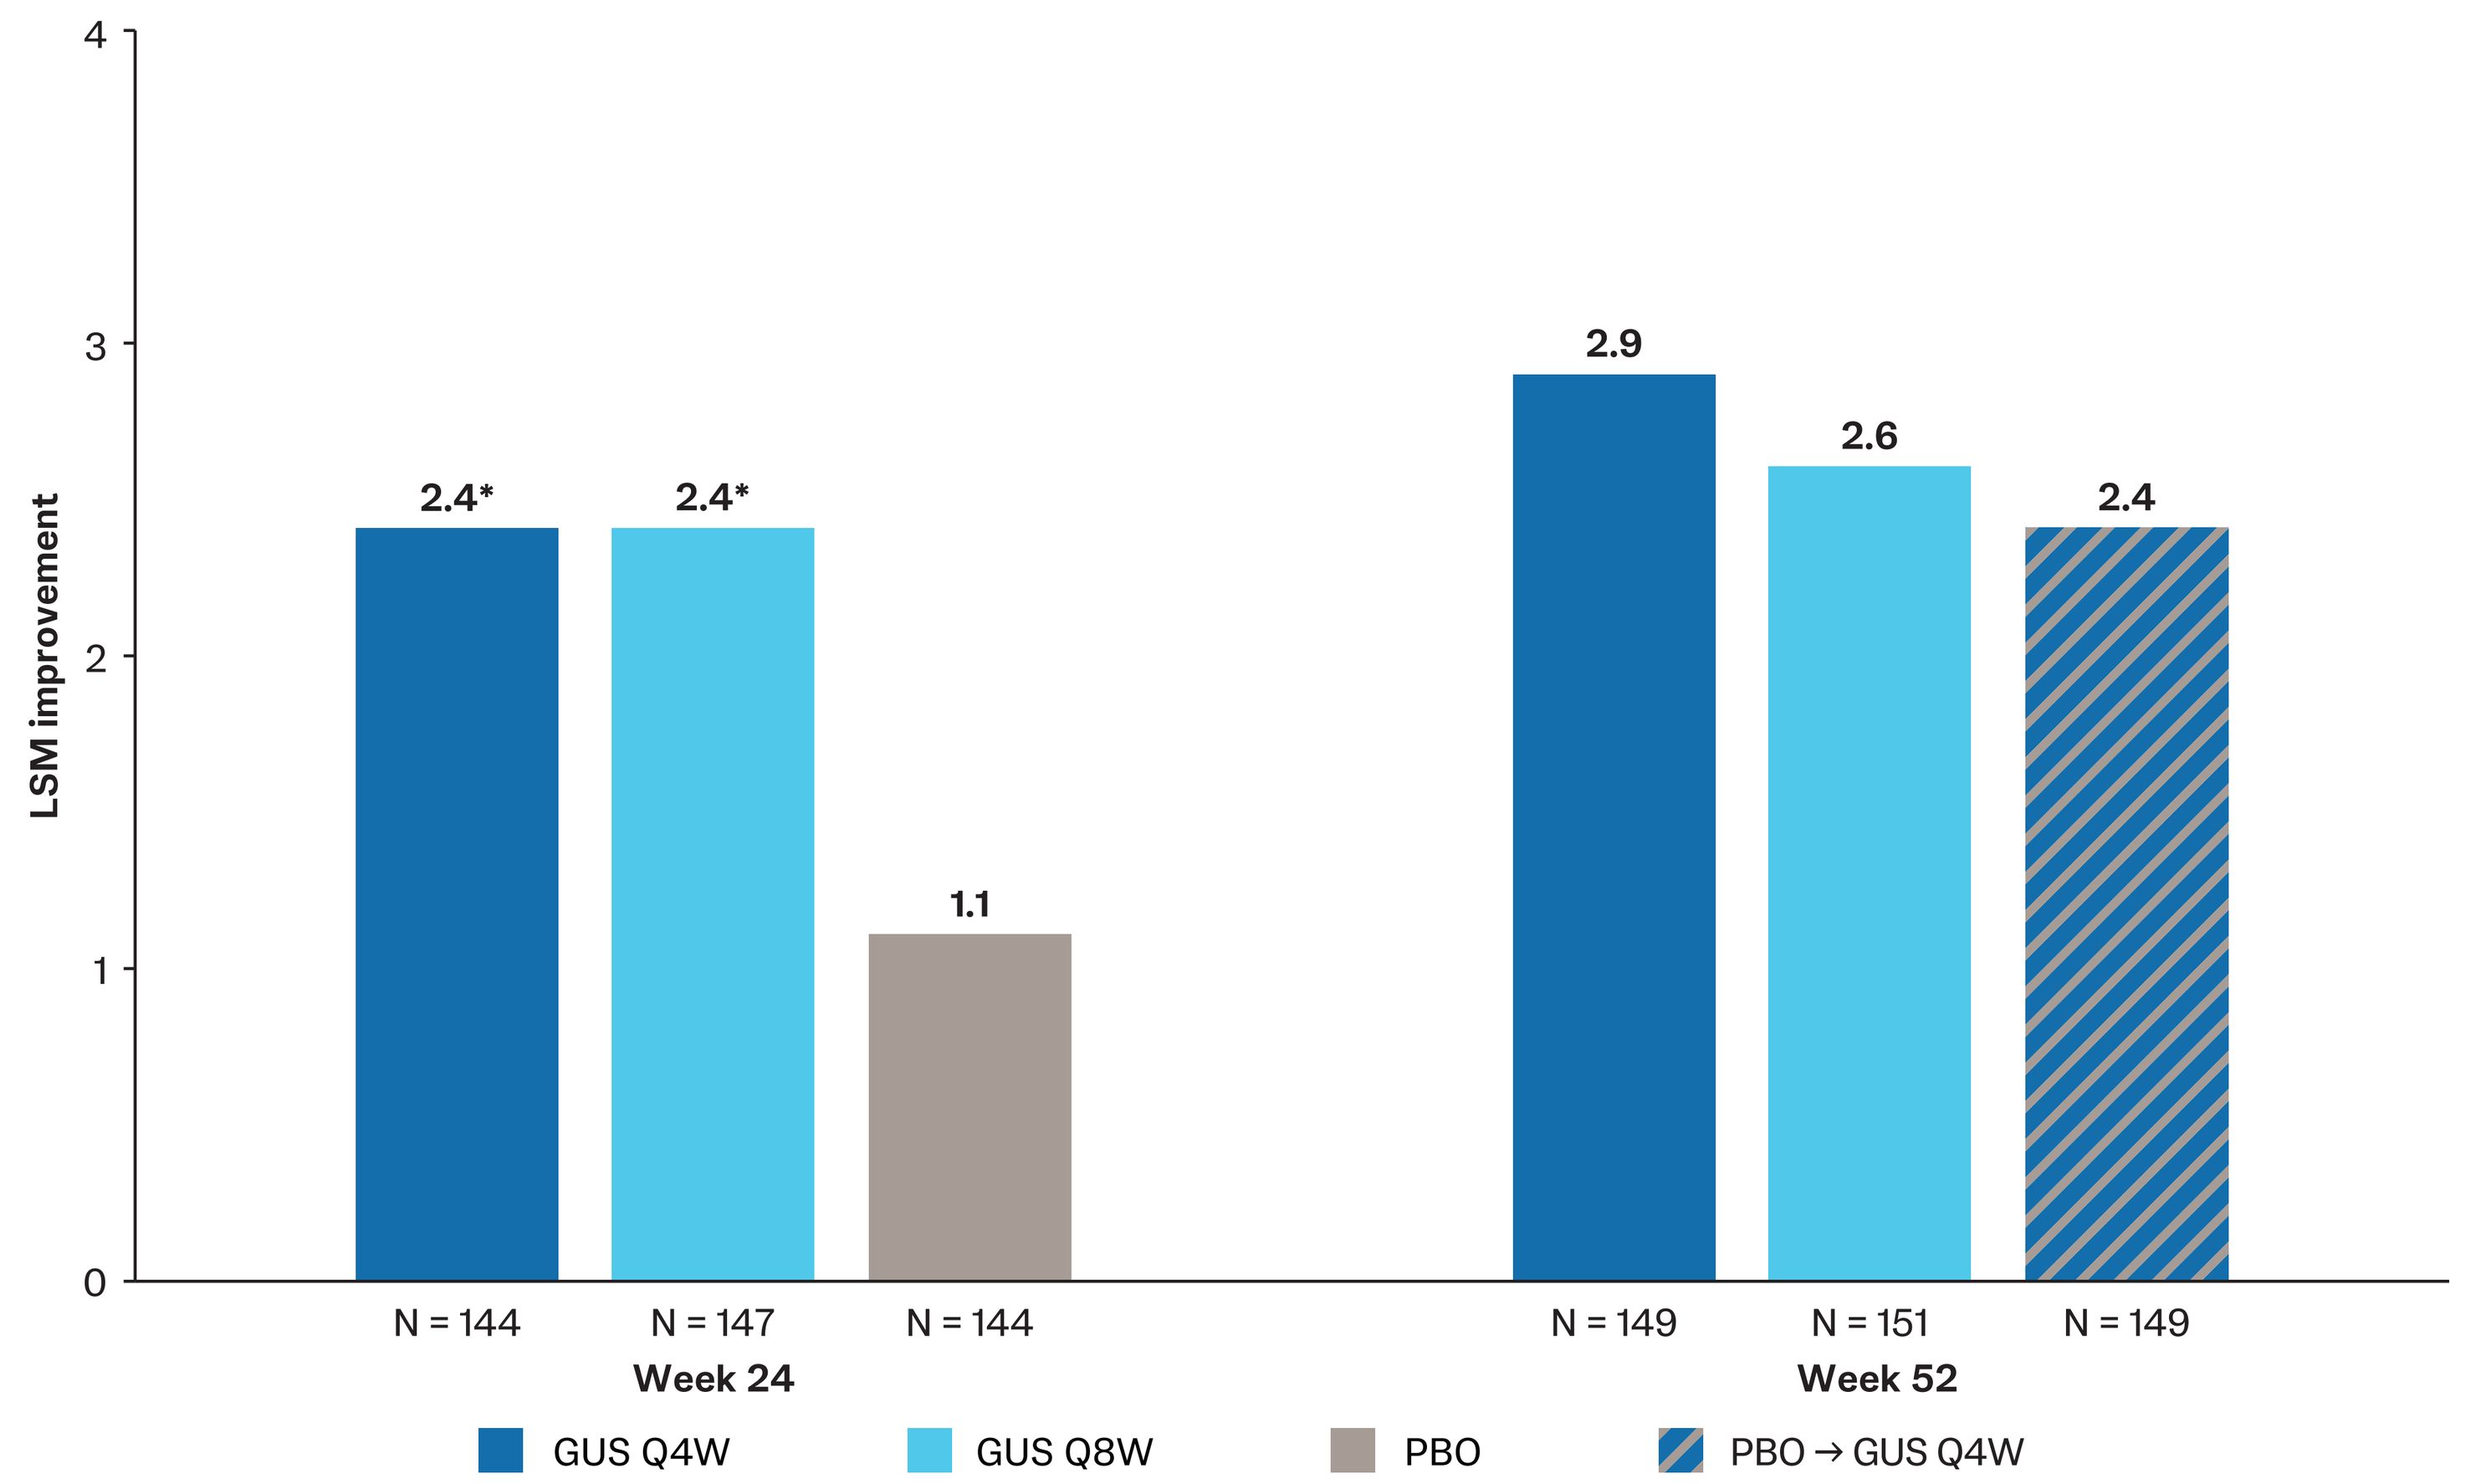

LSM improvement
2.4*
2.4*
1.1
2.9
2.6
2.4
N = 144
N = 147
N = 144
Week 24
N = 149
N = 151
N = 149
Week 52
GUS Q4W
GUS Q8W
PBO
PBO → GUS Q4W

Nominal *p<0.001

LSM and nominal p values for LSM improvements from baseline in PsAID-12 total scores were based on Mixed-Effect Model for Repeated Measures. Participants were considered nonresponders through W24 if they increased dose/initiated csDMARDs or oral corticosteroids or initiated protocol-prohibited PsA therapies and through W52 if they discontinued study agent for any reason other than natural disaster (COVID-19 pandemic)/major disruption (Ukraine-Russia crisis). Data for LSM improvements from baseline in PsAID-12 total scores that were impacted by natural disaster/major disruption were not used; other missing data were accounted for in the analysis model.

Response rates for achieving PsAID-12 MCID and PASS were higher with GUS vs PBO at W24 and numerically increased at W52 with GUS

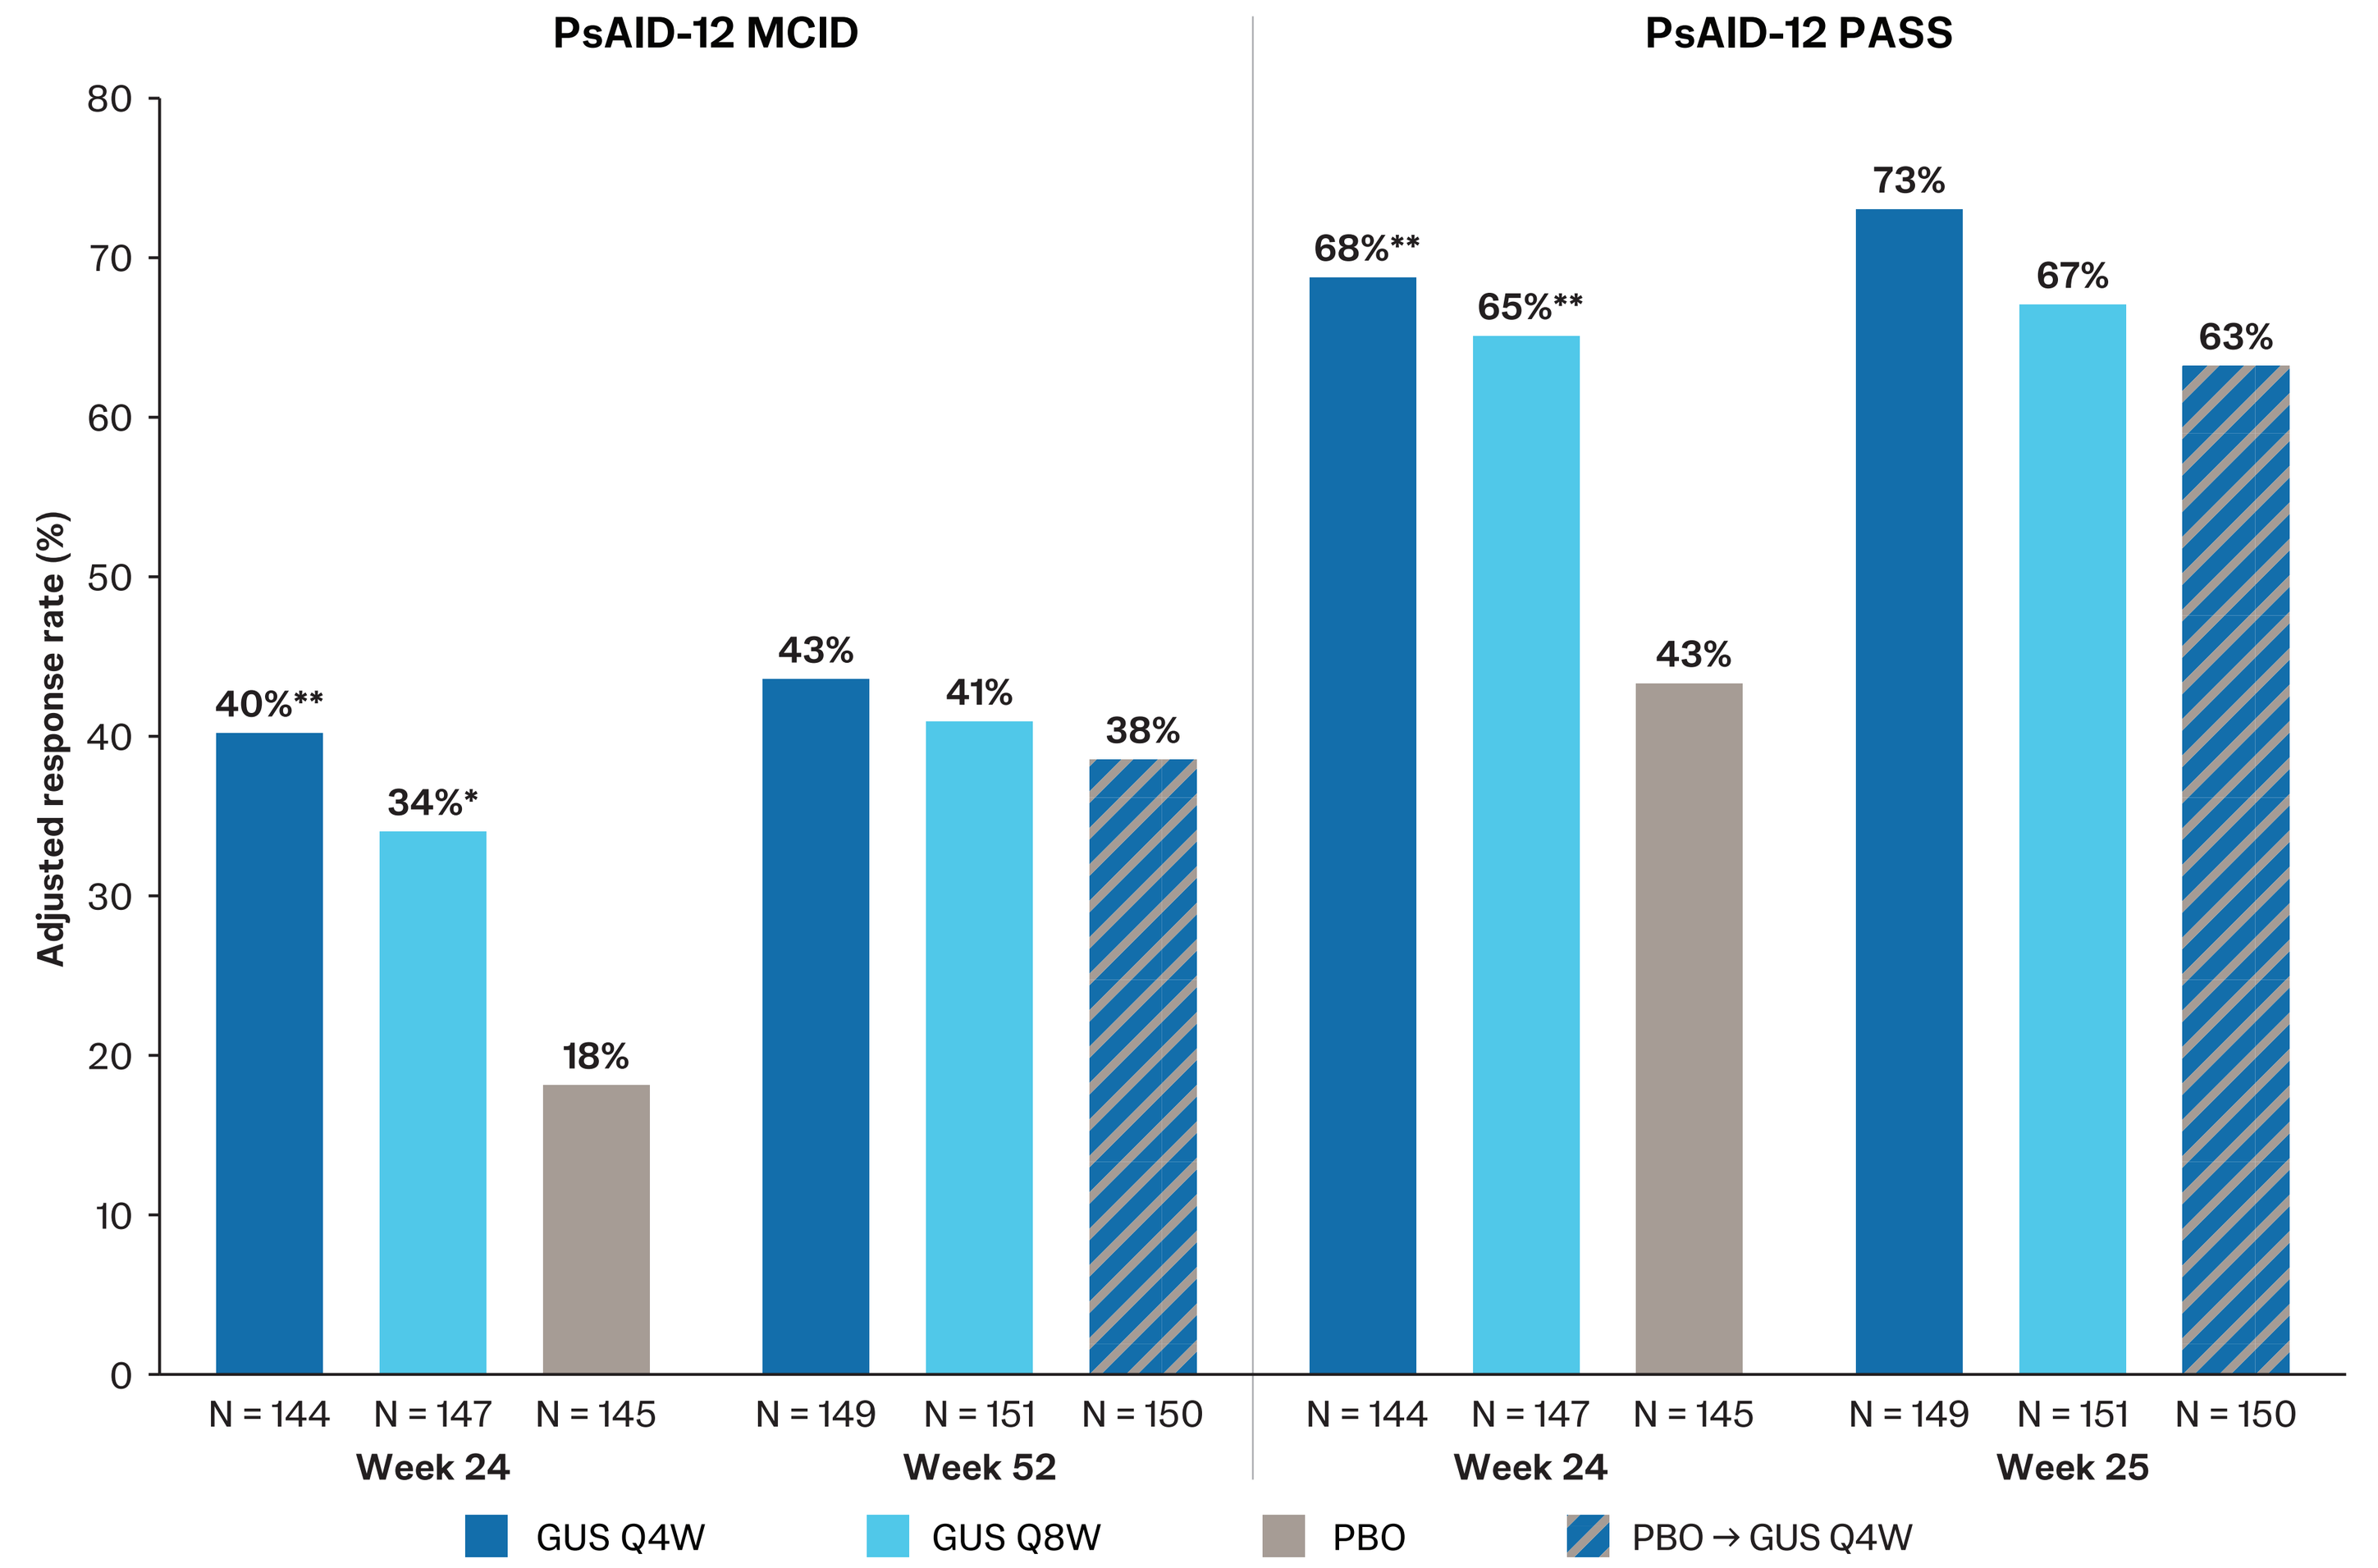

PsAID-12 MCID
PsAID-12 PASS
Adjusted response rate (%)
40%**
34%*
18%
43%
41%
38%
68%**
65%**
43%
73%
67%
63%
N = 144
N = 147
N = 145
Week 24
N = 149
N = 151
N = 150
Week 52
N = 144
N = 147
N = 145
Week 24
N = 149
N = 151
N = 150
Week 25
GUS Q4W
GUS Q8W
PBO
PBO → GUS Q4W

Nominal *p<0.01, **p<0.001

Response rates and nominal p values for proportions of participants achieving PsAID-12 MCID (improvement ≥3) and PASS (score ≤4) were based on a Generalized Linear Mixed Model. Participants were considered nonresponders through W24 if they increased dose/initiated csDMARDs or oral corticosteroids or initiated protocol-prohibited PsA therapies and through W52 if they discontinued study agent for any reason other than natural disaster (COVID-19 pandemic)/major disruption (Ukraine-Russia crisis). Data for proportions of participants achieving PsAID-12 MCID and PASS that were impacted by natural disaster/major disruption were accounted for in the analysis model; other missing data were imputed using nonresponder imputation.

PRESENTED AT: Congress of Clinical Rheumatology-West 2026; September 17-20, 2026; Huntington Beach, CA. Presented by Oyediran Adelakun of Johnson & Johnson, Horsham, PA, USA.

REFERENCES: 1. Singh JA. *Arthritis Rheumatol.* 2019;71:5-32. **2.** Coates LC. *Nat Rev Rheumatol.* 2022;18:465-479. **3.** Sachen KL. *Front Immunol.* 2025;16:1532852. **4.** Ogdie A. *Arthritis Rheumatol.* 2026;78:1674-1686.

ACKNOWLEDGMENTS: Medical writing support was provided by Rozena Varghese, PharmD, CMPP™, of Veristat, Southborough, MA under the direction of the authors in accordance with Good Publication Practice guidelines (*Ann Intern Med.* 2022;175:1298-1304). This presentation was sponsored by Johnson & Johnson. **DISCLOSURES: AO:** Consultant for AbbVie, Amgen, Bristol Myers Squibb, CorEvitas, Eli Lilly, Gilead, GlaxoSmithKline, Johnson & Johnson, Merck, Novartis, Oruka, Pfizer, Spyre, Takeda, and UCB; received advisory board fees from AbbVie, Amgen, Bristol Myers Squibb, Eli Lilly, Gilead, GlaxoSmithKline, Johnson & Johnson, Novartis, Pfizer, and UCB; received grants from AbbVie, Pfizer, and Novartis (all to University of Pennsylvania) and Amgen (to Forward/NDB); and received other funding from NIAMS, Rheumatology Research Foundation, National Psoriasis Foundation, and University of Pennsylvania. **ABG:** Received research/educational grants from Bristol Myers Squibb, Johnson & Johnson, Moonlake, and UCB, (all paid to Mount Sinai School of Medicine until May 1, 2025); at UT Southwestern, Dr. Gottlieb is a subinvestigator on studies sponsored by Bristol Myers Squibb and Johnson & Johnson and principal investigator on a study sponsored by Bristol Myers Squibb; and Dr. Gottlieb received honoraria as an advisory board member and consultant for AbbVie, Amgen, Biogen, Bristol Myers Squibb, Eli Lilly, Johnson & Johnson, Novartis, Oruka, Sanofi, SunPharma, Takeda, Teva, and UCB. **PJM:** Received consulting fees from AbbVie, Amgen, Bristol Myers Squibb, Century, Cullinan, Eli Lilly, Inmagene, Johnson & Johnson, Merck, Moonlake, Novartis, Pfizer, Spyre, Sun Pharma, Takeda, and UCB; received grants from AbbVie, Amgen, Bristol Myers Squibb, Eli Lilly, Johnson & Johnson, Moonlake, Novartis, Sana, Takeda, and UCB; and received speaker fees from AbbVie, Amgen, Eli Lilly, Johnson & Johnson, Novartis, and UCB. **JFM:** Consultant and/or investigator for AbbVie, Amgen, AstraZeneca, Biogen, Boehringer Ingelheim, Bristol Myers Squibb, Dermavant, Eli Lilly, Galderma, Johnson & Johnson, Moonlake, Novartis, Oruka, Pfizer, Regeneron, Sanofi, Sun Pharma, and UCB. **EA, SDC, FI,** and **KPL:** Employees of Johnson & Johnson; may own stock/stock options in Johnson & Johnson. **YW:** Employee of IQVIA providing statistical support (funded by Johnson & Johnson). **CTR:** Consultant for AbbVie, Eli Lilly, Johnson & Johnson, Novartis, and UCB; and received grant/research support from Johnson & Johnson, Novartis, and UCB. **JUS:** Received grant/research support from Johnson & Johnson and Pfizer; and received consulting fees from Bristol Myers Squibb, Johnson & Johnson, Novartis, Pfizer, and UCB. **PRESENTER DISCLOSURES: OA:** Employee of Johnson & Johnson; owns stock/stock options in Johnson & Johnson.

This material is distributed for scientific purposes by Johnson & Johnson Innovative Medicine and is not for promotional use.