Nipocalimab in SLE: First-in-Class Efficacy and Safety Results Demonstrating Proof of Concept for FcRn Blockade From the Phase 2 JASMINE-SLE Study

Richard A Furie,[1] Ronald van Vollenhoven,[2] Rafał Wojciechowski,[3] Eric Morand,[4] Guillermo J. Pons-Estel,[5] George A. Karpouzas,[6] Bart van Hartingsveldt,[7] Robert W. Hoffman,[8] Terence Rooney,[8] Sheng Gao,[8] Robert Gordon,[8] Kim Hung Lo,[8] Thanh Nguyen,[9] Jocelyn H. Leu,[8] Federico Zazzetti,[8] Stanley J. Marciniak,[8] Steven Leonardo,[8] Keying Ma,[8] Kathy Sivils,[8] Leonard Dragone,[10] Loqmane Seridi,[8] Linda Okonkwo,[11] Erika Noss,[8] Fang Liu-Walsh,[8] Michelle Petri[12]

[1]Division of Rheumatology, Northwell, Great Neck, NY, USA; [2]University of Amsterdam, Amsterdam, Noord-Holland, The Netherlands; [3]Department of Rheumatology and Systemic Connective Tissue Diseases, University Hospital No. 2, Bydgoszcz, Poland; [4]Centre for Inflammatory Diseases, Monash University, Melbourne, VIC, Australia; [5]Centro Regional de Enfermedades Autoinmunes y Reumáticas (GO-CREAR), Grupo Oroño, Rosario, Argentina; [6]Harbor-UCLA Medical Center, Division of Rheumatology and the Lundquist Institute for Biomedical Innovation, Torrance, CA, USA; [7]Johnson & Johnson, Leiden, Netherlands; [8]Johnson & Johnson, Spring House, PA, USA; [9]Johnson & Johnson, Horsham, PA, USA; [10]Johnson & Johnson, San Francisco, CA, USA; [11]Johnson & Johnson, Raritan, NJ, USA; [12]Johns Hopkins University School of Medicine, Baltimore, Maryland, USA

Scan the QR code.
The QR code is intended to provide scientific information for individual reference, and the information should not be altered or reproduced in any way.

Key Takeaways

- **JASMINE-SLE, the first study of an FcRn blocker in SLE, demonstrated that:**
 - **Nipocalimab 15 mg/kg IV Q2W treatment led to significant improvement over placebo in SRI-4 composite response at Week 24 and improved SRI-4 composite and LLDAS responses at Week 52**
 - **Nipocalimab 15 mg/kg treatment led to sustained oral GC reduction**
 - **Greater treatment effects were observed in populations of aAb-positive, aAb-high, and IFN-high participants**
 - **Nipocalimab was well tolerated among participants**
- **These results establish proof of concept for FcRn blockade with nipocalimab in SLE and support the evaluation of nipocalimab in an ongoing phase 3 trial (GARDENIA; NCT07438496)**

Background

- Systemic lupus erythematosus (SLE) is a chronic autoantibody-driven disease in which IgG autoantibodies (aAbs) contribute to systemic inflammation that leads to tissue injury and organ damage through direct cytotoxicity (via cellular binding, ADCC, ADCP, and CDCC) and the formation of circulating immune complexes (CICs)[1,2]
- The neonatal Fc receptor (FcRn) maintains circulating IgG levels through recycling and transcytosis, and is expressed across multiple cell types including monocytes and endothelial cells[3–5]
- Nipocalimab is a high-affinity, fully human, monoclonal antibody that selectively binds the neonatal FcRn, thereby blocking the binding of endogenous IgG to FcRn and promoting lysosomal degradation of circulating IgG, including pathogenic IgG aAbs, and CICs, while preserving immune cell functions[6–8]
 - Nipocalimab is approved for generalized myasthenia gravis (gMG) in the US, Brazil, Japan, Europe and China[9]

Objective

To evaluate the efficacy, safety, and pharmacodynamics of nipocalimab in adults with active SLE through a multicenter, randomized, double-blind, placebo-controlled, parallel-group study (JASMINE-SLE)

Methods

Key eligibility criteria
- ✓ Adults with active SLE (SLICC 2012) for ≥6 months
- ✓ Received standard of care (antimalarials, oral GCs, and protocol-permitted immunomodulators)
- ✓ Positive for ANA, anti-dsDNA, and/or anti-Smith
- ✓ 1 BILAG A and/or 2 BILAG B scores
- ✓ SLEDAI-2K score ≥6

Primary endpoint[a]
- ✓ % participants achieving SRI-4 composite response at Week 24

Key secondary/exploratory endpoints[b]
- ✓ % participants achieving SRI-4 composite response at Week 52
- ✓ % participants achieving LLDAS response at Week 52

Oral GC taper
- ✓ **Mandatory taper:** Weeks 6 to 16 to a target of 7.5 mg/day and maintained through Week 24[c]
- ✓ **Recommended taper:** Weeks 24 to 40 to a target of 7.5 mg/day and maintained through Week 52

JASMINE — Study Design

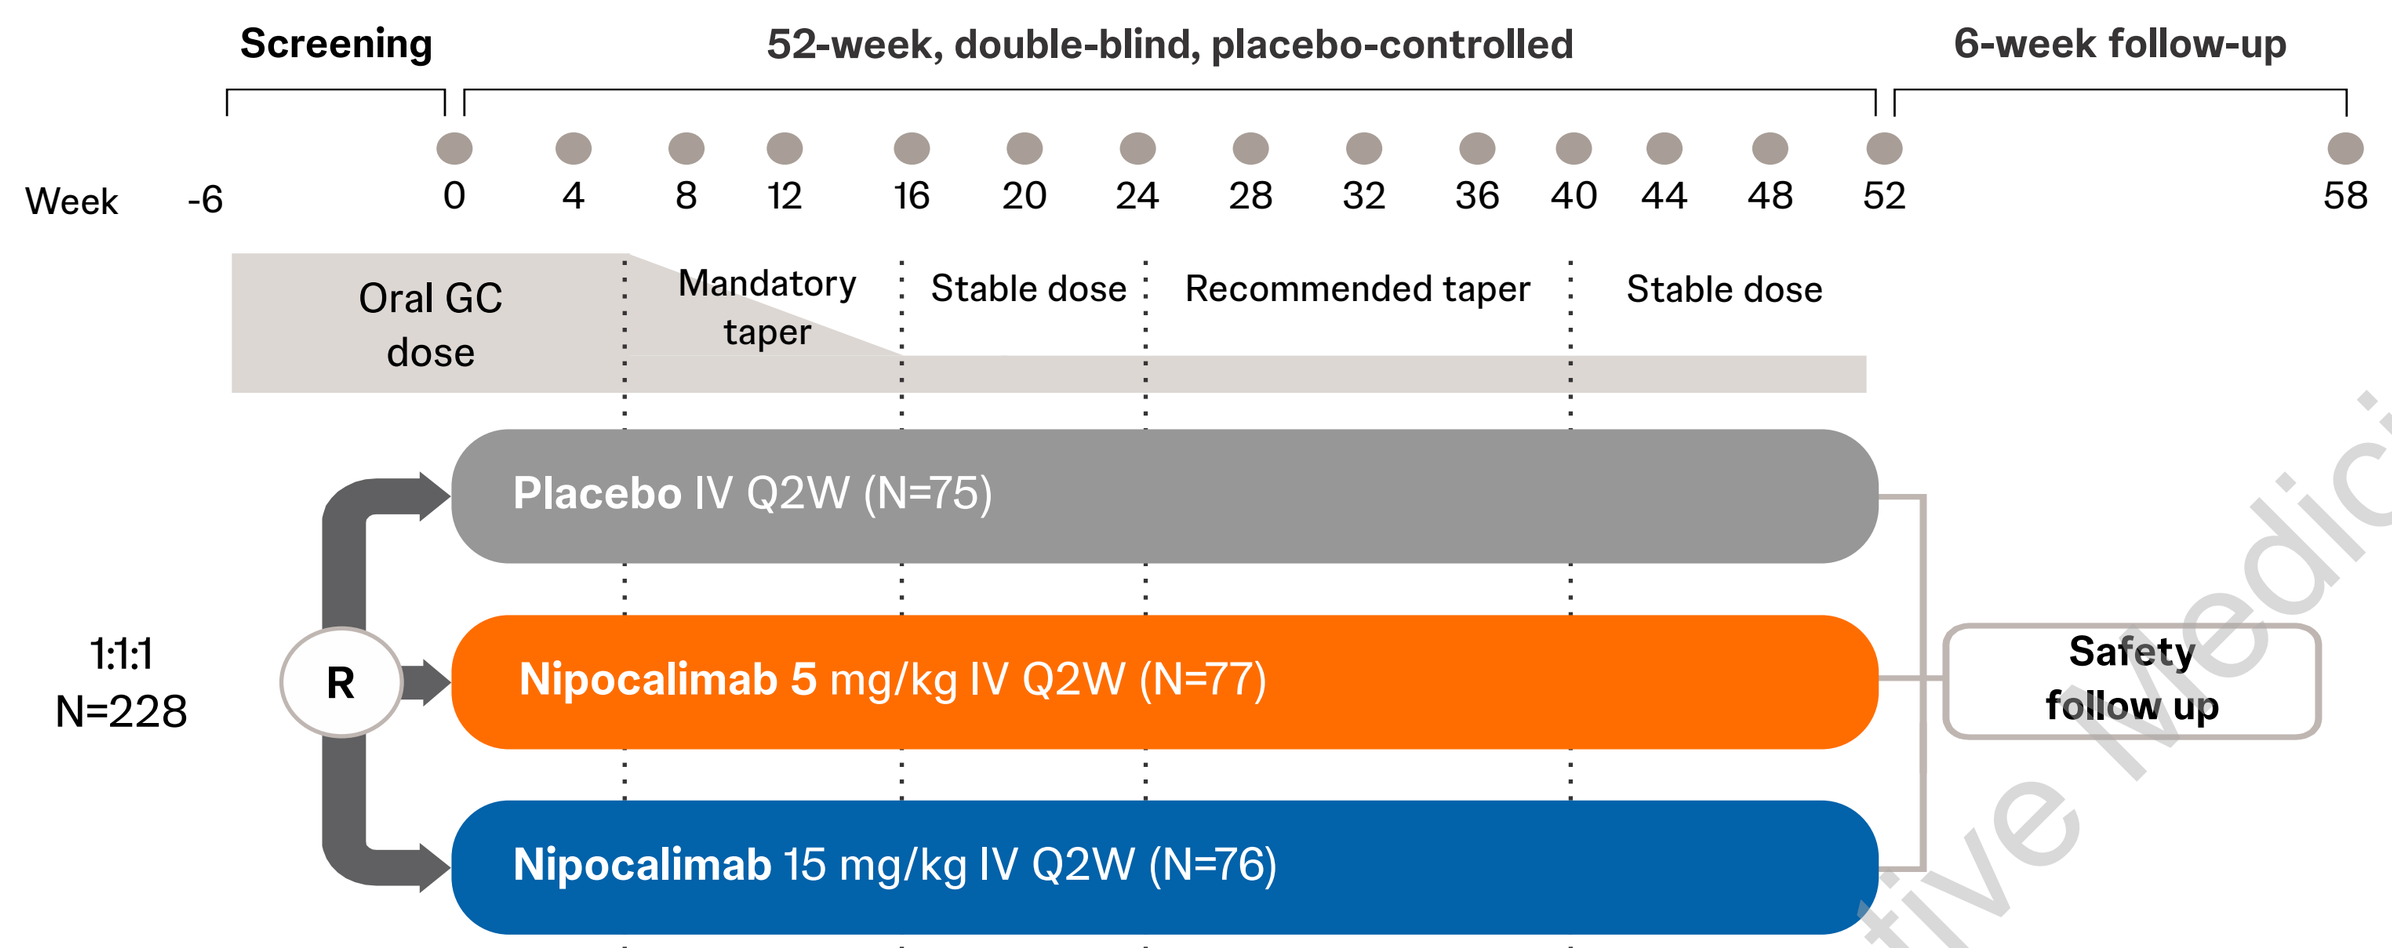
Screening
52-week, double-blind, placebo-controlled
6-week follow-up
Week -6 0 4 8 12 16 20 24 28 32 36 40 44 48 52 58
Oral GC dose
Mandatory taper
Stable dose
Recommended taper
Stable dose
1:1:1
N=228
R
Placebo IV Q2W (N=75)
Nipocalimab 5 mg/kg IV Q2W (N=77)
Nipocalimab 15 mg/kg IV Q2W (N=76)
Safety follow up

aAb-positive (79%)
- Positive for ≥1:
 - Anti-dsDNA
 - Anti-Smith
 - ANA with anti-RNP, anti-Ro, or historical anti-dsDNA
- Positive for a range of additional autoantibodies based on extended profiling

aAb-high (15%)
- Identified using machine learning through unsupervised clustering of baseline aAb levels
- Consisted of patients with (all present):
 - Increased CICs
 - High IFN signature
 - Low C3

IFN-high (66%)
- Defined using a peripheral blood RNA signature[10]

[a]Only the primary endpoint was controlled for multiplicity, with a prespecified 2-sided alpha level of 0.10. [b]All other efficacy analyses were not adjusted for multiplicity, with nominal P values reported. [c]Participants on ≥10 mg/day oral GCs were required to taper; tapering could be paused/increased for disease worsening or safety concerns. FAS included all randomized participants who received ≥1 dose of any study intervention (for safety analyses and primary endpoint analysis). mFAS included all randomized participants who received ≥1 dose of any study intervention, excluding 7 participants from one study site due to noncompliance with Good Clinical Practice identified between Week 24 and Week 52, for Week 52 efficacy analyses.

ANA=antinuclear antibody, BILAG=British Isles Lupus Assessment Group Scale, dsDNA=double-stranded deoxyribonucleic acid, FAS=full analysis set, GC=glucocorticoid, IV=intravenous, LLDAS=Lupus Low Disease Activity State, mFAS=modified full analysis set, Q2W=every 2 weeks, R=randomization, SLE=systemic lupus erythematosus, SLEDAI-2K=Systemic Lupus Erythematosus Disease Activity Index 2000, SLICC=Systemic Lupus International Collaborating Clinics criteria, SRI-4=Systemic Lupus Erythematosus Responder Index.

aAb=autoantibody, ANA=antinuclear antibody, C3=complement component 3, CIC=circulating immune complex, dsDNA=double-stranded deoxyribonucleic acid, IFN=interferon, RNP=ribonucleoprotein.

Results

Demographic and baseline clinical characteristics were comparable across treatment groups

Baseline characteristics[a]	Placebo (N=73)	Nipocalimab 5 mg/kg IV Q2W (N=74)	Nipocalimab 15 mg/kg IV Q2W (N=74)	Total (N=221)
Demographics				
Age, years, mean (SD)	45.5 (11.3)	40.9 (11.9)	44.0 (11.9)	43.4 (11.8)
Female, n (%)	69 (94.5)	70 (94.6)	70 (94.6)	209 (94.6)
White, n (%)	50 (68.5)	50 (67.6)	44 (59.5)	144 (65.2)
Disease characteristics, mean (SD)				
SLE duration, years	9.3 (8.0)	8.1 (6.0)	10.8 (9.6)	9.4 (8.0)
SLEDAI-2K score (0–105)	**10.4 (3.0)**	**10.0 (3.2)**	**9.8 (2.5)**	**10.1 (2.9)**
PGA score (0–3)	2.0 (0.3)	1.9 (0.3)	2.0 (0.3)	2.0 (0.3)
BILAG, n (%)				
≥1 BILAG A	41 (56.2)	40 (54.1)	50 (67.6)	131 (59.3)
≥2 BILAG B	31 (42.5)	32 (43.2)	24 (32.4)	87 (39.4)
≥1 BILAG A or ≥2 BILAG B	72 (98.6)	71 (95.9)	72 (97.3)	215 (97.3)
CLASI activity score (0–70)	7.1 (4.5)	8.2 (7.2)	7.6 (6.7)	7.6 (6.2)
Number of active joints (0–62)[b]	8.0 (4.6)	8.6 (4.5)	10.1 (7.1)	8.9 (5.6)
Total IgG, g/L	14.2 (4.8)	15.2 (6.8)	14.4 (5.4)	14.6 (5.7)
Anti-dsDNA (positive ≥75), IU/mL	**167.0 (467.2)**	**155.7 (354.1)**	**225.5 (663.2)**	**182.8 (509.8)**
C3 (0.9–1.8), g/L	1.1 (0.3)	1.1 (0.3)	1.1 (0.3)	1.1 (0.3)
CIC,[c] µg Eq/mL	11.4 (7.9)	12.7 (9.2)	11.9 (8.1)	12.0 (8.4)
Concomitant medications, n (%)				
Antimalarials	62 (84.9)	61 (82.4)	64 (86.5)	187 (84.6)
Oral GCs	**46 (63.0)**	**59 (79.7)**	**52 (70.3)**	**157 (71.0)**
Immunomodulators	**38 (52.1)**	**33 (44.6)**	**35 (47.3)**	**106 (48.0)**

[a]Based on the mFAS. Sample sizes for each parameter reflect nonmissing values. [b]Active joints are joints that are painful (as reported by participants) and tender and swollen (on physical exam as determined by the joint assessor). [c]Based on the mFAS with available baseline CIC values (placebo n=72, nipocalimab 5 mg/kg n=71, nipocalimab 15 mg/kg n=72).

BILAG=British Isles Lupus Assessment Group Scale, C3=complement component 3, CIC=circulating immune complex, CLASI=Cutaneous Lupus Erythematosus Disease Area and Severity Index, dsDNA=double-stranded deoxyribonucleic acid, Eq=equivalent, GC=glucocorticoid, IgG=immunoglobulin G, IV=intravenous, mFAS=modified full analysis set, PGA=Physician's Global Assessment of Disease Activity, Q2W=every 2 weeks, SD=standard deviation, SLE=systemic lupus erythematosus, SLEDAI-2K=Systemic Lupus Erythematosus Disease Activity Index 2000.

SRI-4 composite response at Week 24 was met for nipocalimab 15 mg/kg versus placebo (53.5% vs 46.7% P=0.081[a]) but not for nipocalimab 5 mg/kg versus placebo

- Among participants eligible for oral GC taper, a higher proportion of participants treated with 15 mg/kg nipocalimab achieved sustained oral GC taper and achieved SRI-4 composite response with sustained taper at Week 24

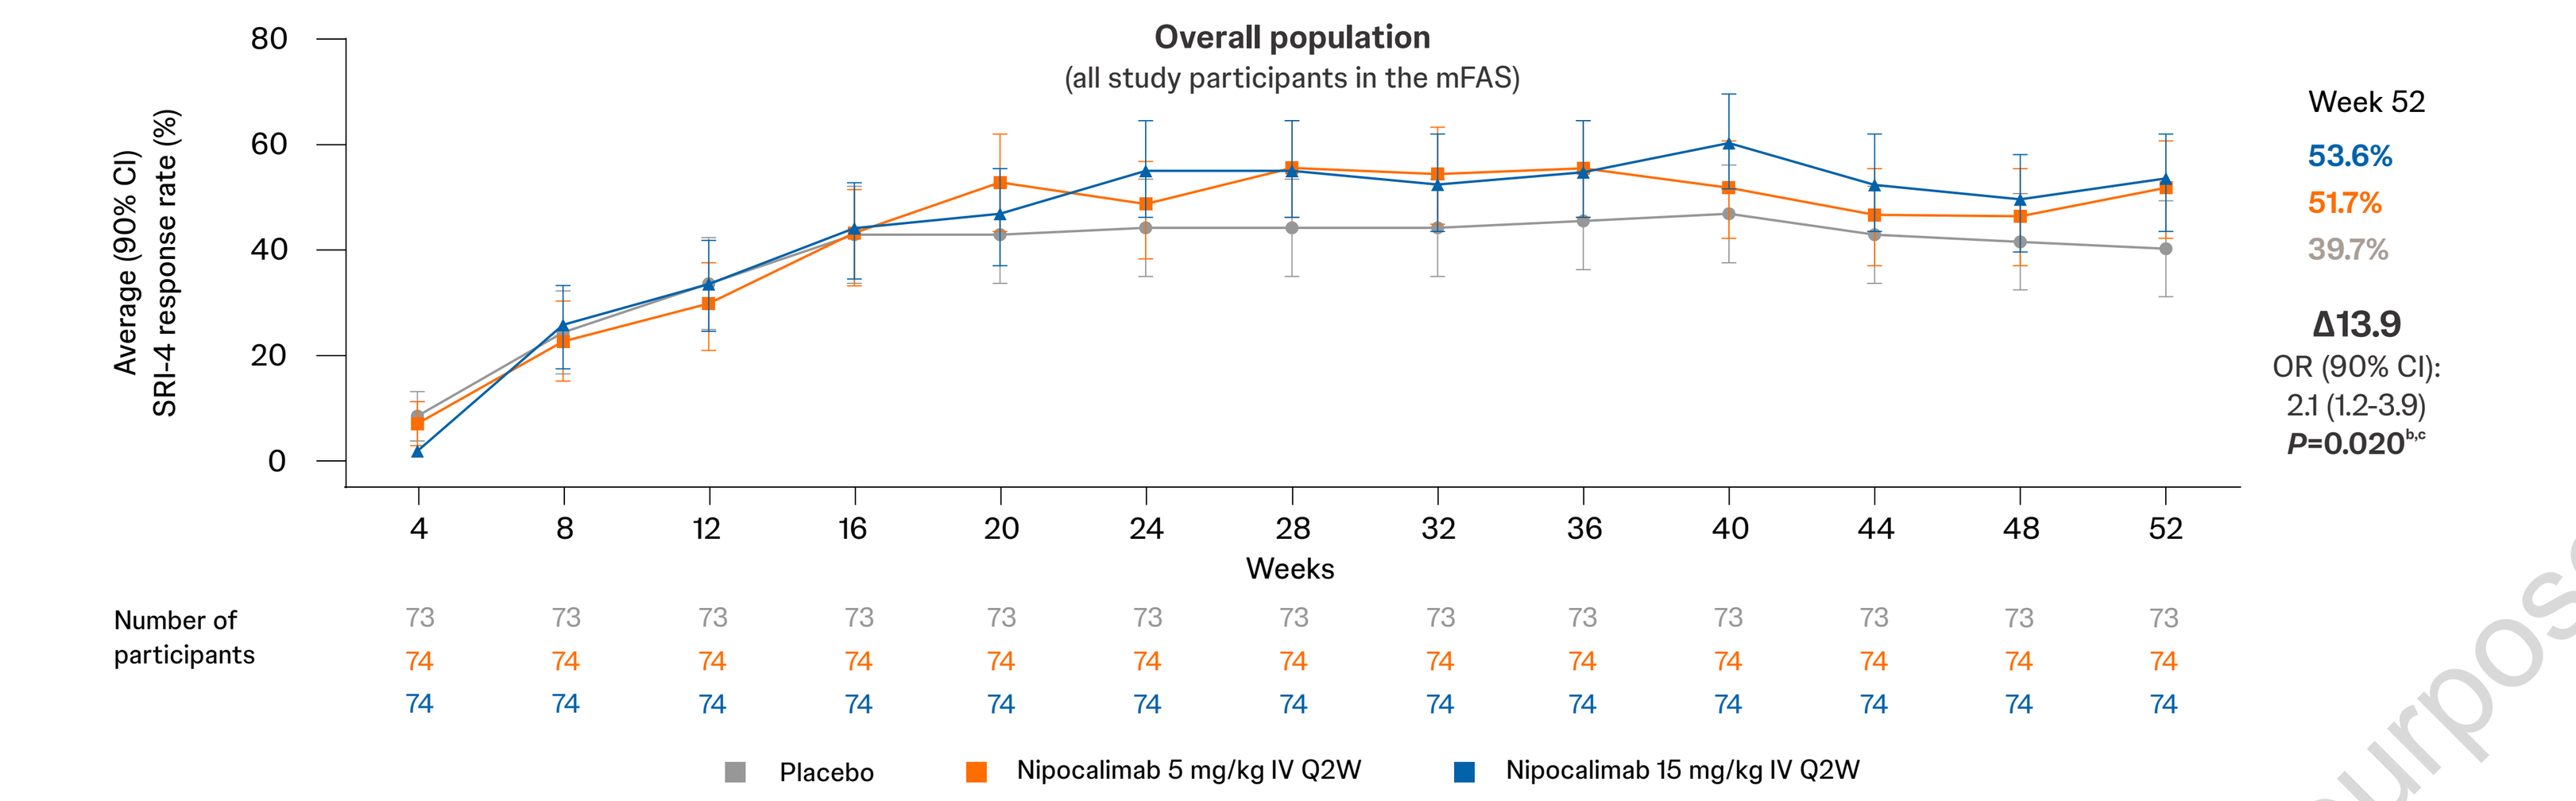
Overall population
(all study participants in the mFAS)
Average (90% CI) SRI-4 response rate (%)
Weeks
Number of participants
Week 52
53.6%
51.7%
39.7%
Δ13.9
OR (90% CI): 2.1 (1.2-3.9)
P=0.020[b,c]
Placebo
Nipocalimab 5 mg/kg IV Q2W
Nipocalimab 15 mg/kg IV Q2W

[a]Only the primary endpoint was controlled for multiplicity, with a prespecified 2-sided alpha level of 0.10 across doses, appropriate for a phase 2, proof-of-concept study. FAS included all randomized participants who received ≥1 dose of any study intervention (for safety analyses and primary endpoint analysis). mFAS included all randomized participants who received ≥1 dose of any study intervention, excluding 7 participants from one study site due to noncompliance with Good Clinical Practice identified between Week 24 and Week 52, for Week 52 efficacy analyses. [b]Analysis based on average response rates with imputed data. [c]The P value was nominal.

aAb=autoantibody, CI=confidence interval, FAS=full analysis set, GC=glucocorticoid, IV=intravenous, mFAS=modified full analysis set, OR=odds ratio, Q2W=every 2 weeks, SRI-4=Systemic Lupus Erythematosus Responder Index.

Nipocalimab improved Week 52 SRI-4 composite response rates compared with placebo, with greater response rates in predefined biomarker-based populations

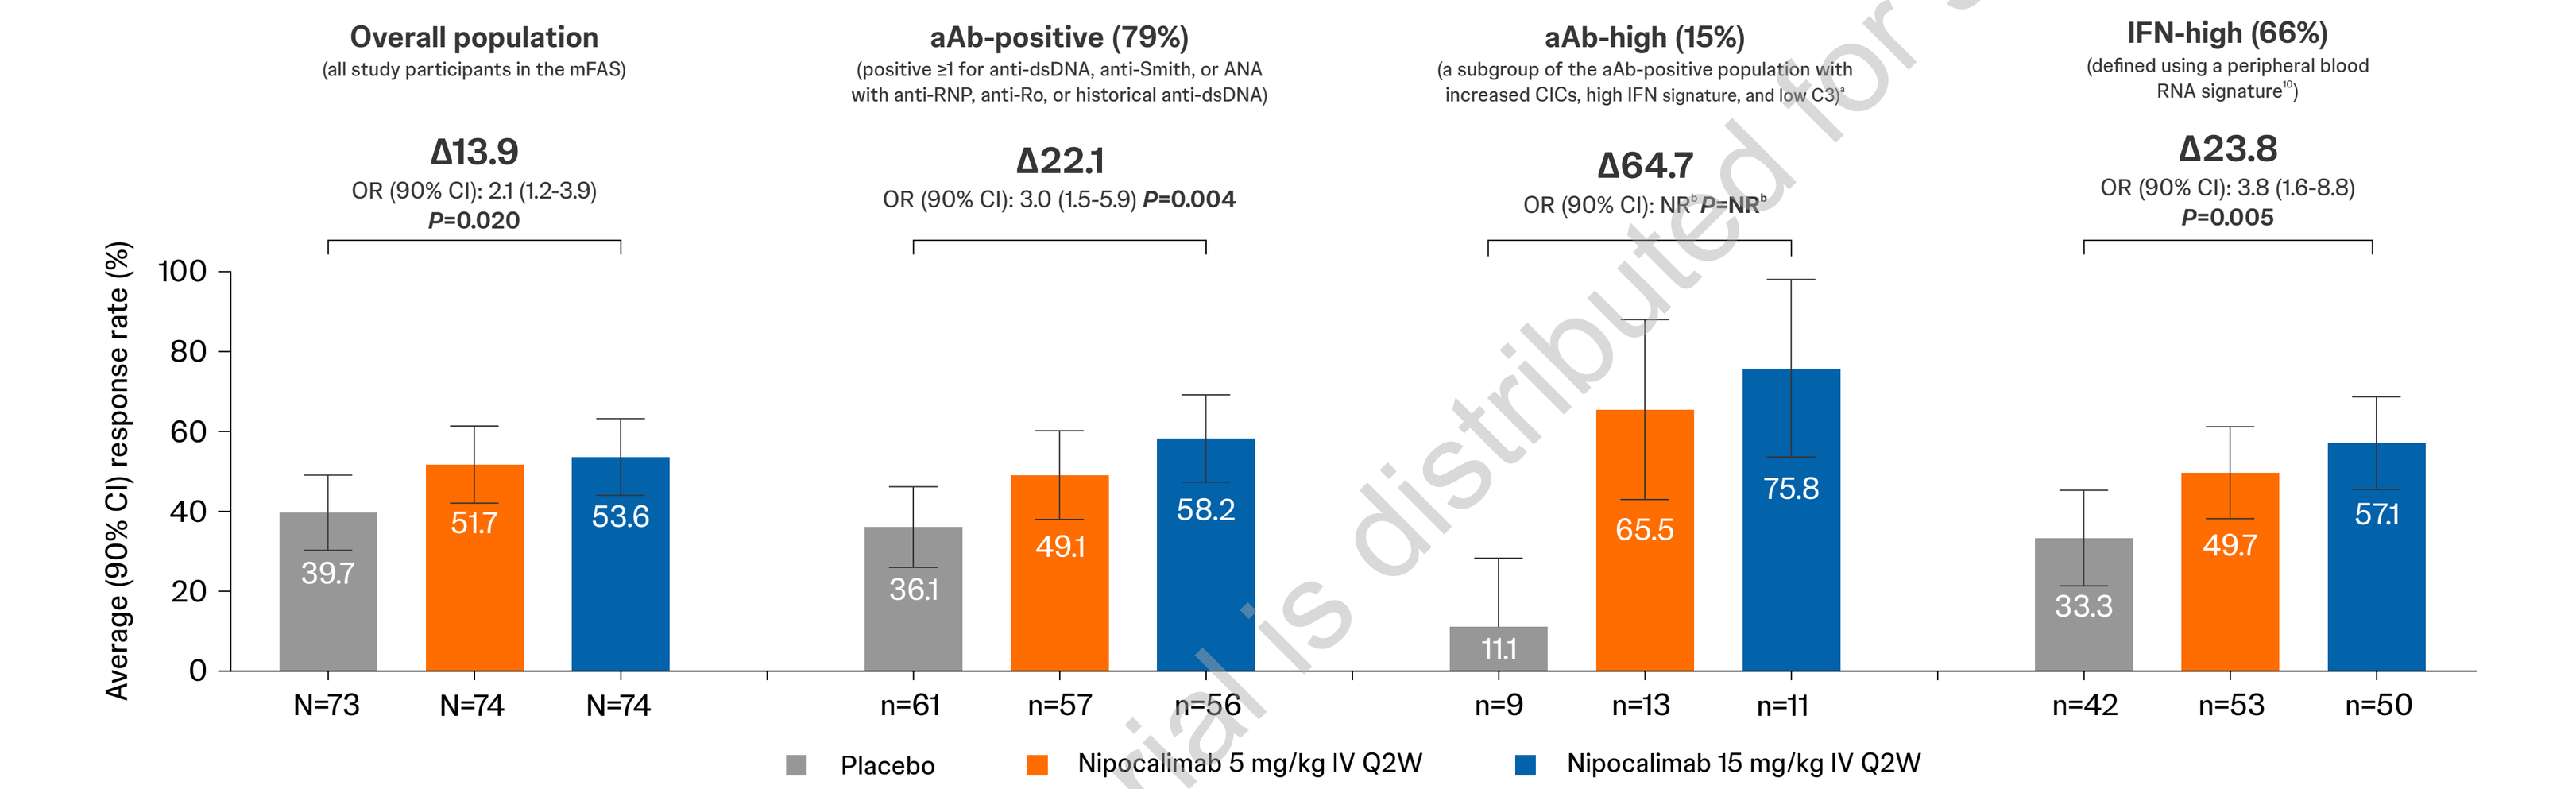
Overall population
(all study participants in the mFAS)
Δ13.9
OR (90% CI): 2.1 (1.2-3.9) P=0.020
aAb-positive (79%)
(positive ≥1 for anti-dsDNA, anti-Smith, or ANA with anti-RNP, anti-Ro, or historical anti-dsDNA)
Δ22.1
OR (90% CI): 3.0 (1.5-5.9) P=0.004
aAb-high (15%)
(a subgroup of the aAb-positive population with increased CICs, high IFN signature, and low C3)[a]
Δ64.7
OR (90% CI): NR[b] P=NR[b]
IFN-high (66%)
(defined using a peripheral blood RNA signature[10])
Δ23.8
OR (90% CI): 3.8 (1.6-8.8) P=0.005
Average (90% CI) response rate (%)
39.7 51.7 53.6
N=73 N=74 N=74
36.1 49.1 58.2
n=61 n=57 n=56
11.1 65.5 75.8
n=9 n=13 n=11
33.3 49.7 57.1
n=42 n=53 n=50
Placebo
Nipocalimab 5 mg/kg IV Q2W
Nipocalimab 15 mg/kg IV Q2W

Analyses based on the mFAS with imputed data. All P values reported here are nominal. [a]Identified through unsupervised consensus clustering based on semiquantitative baseline aAb intensities. [b]Due to small sample sizes and large variability, the OR and P value are NR.

aAb=autoantibody, ANA=antinuclear antibody, C3=complement component 3, CI=confidence interval, CIC=circulating immune complex, dsDNA=double-stranded deoxyribonucleic acid, IFN=interferon, IV=intravenous, mFAS=modified full analysis set, NR=not reported, OR=odds ratio, Q2W=every 2 weeks, RNP=ribonucleoprotein, SRI-4=Systemic Lupus Erythematosus Responder Index.

Nipocalimab improved Week 52 LLDAS response rates compared with placebo, with greater response rates in predefined biomarker-based populations

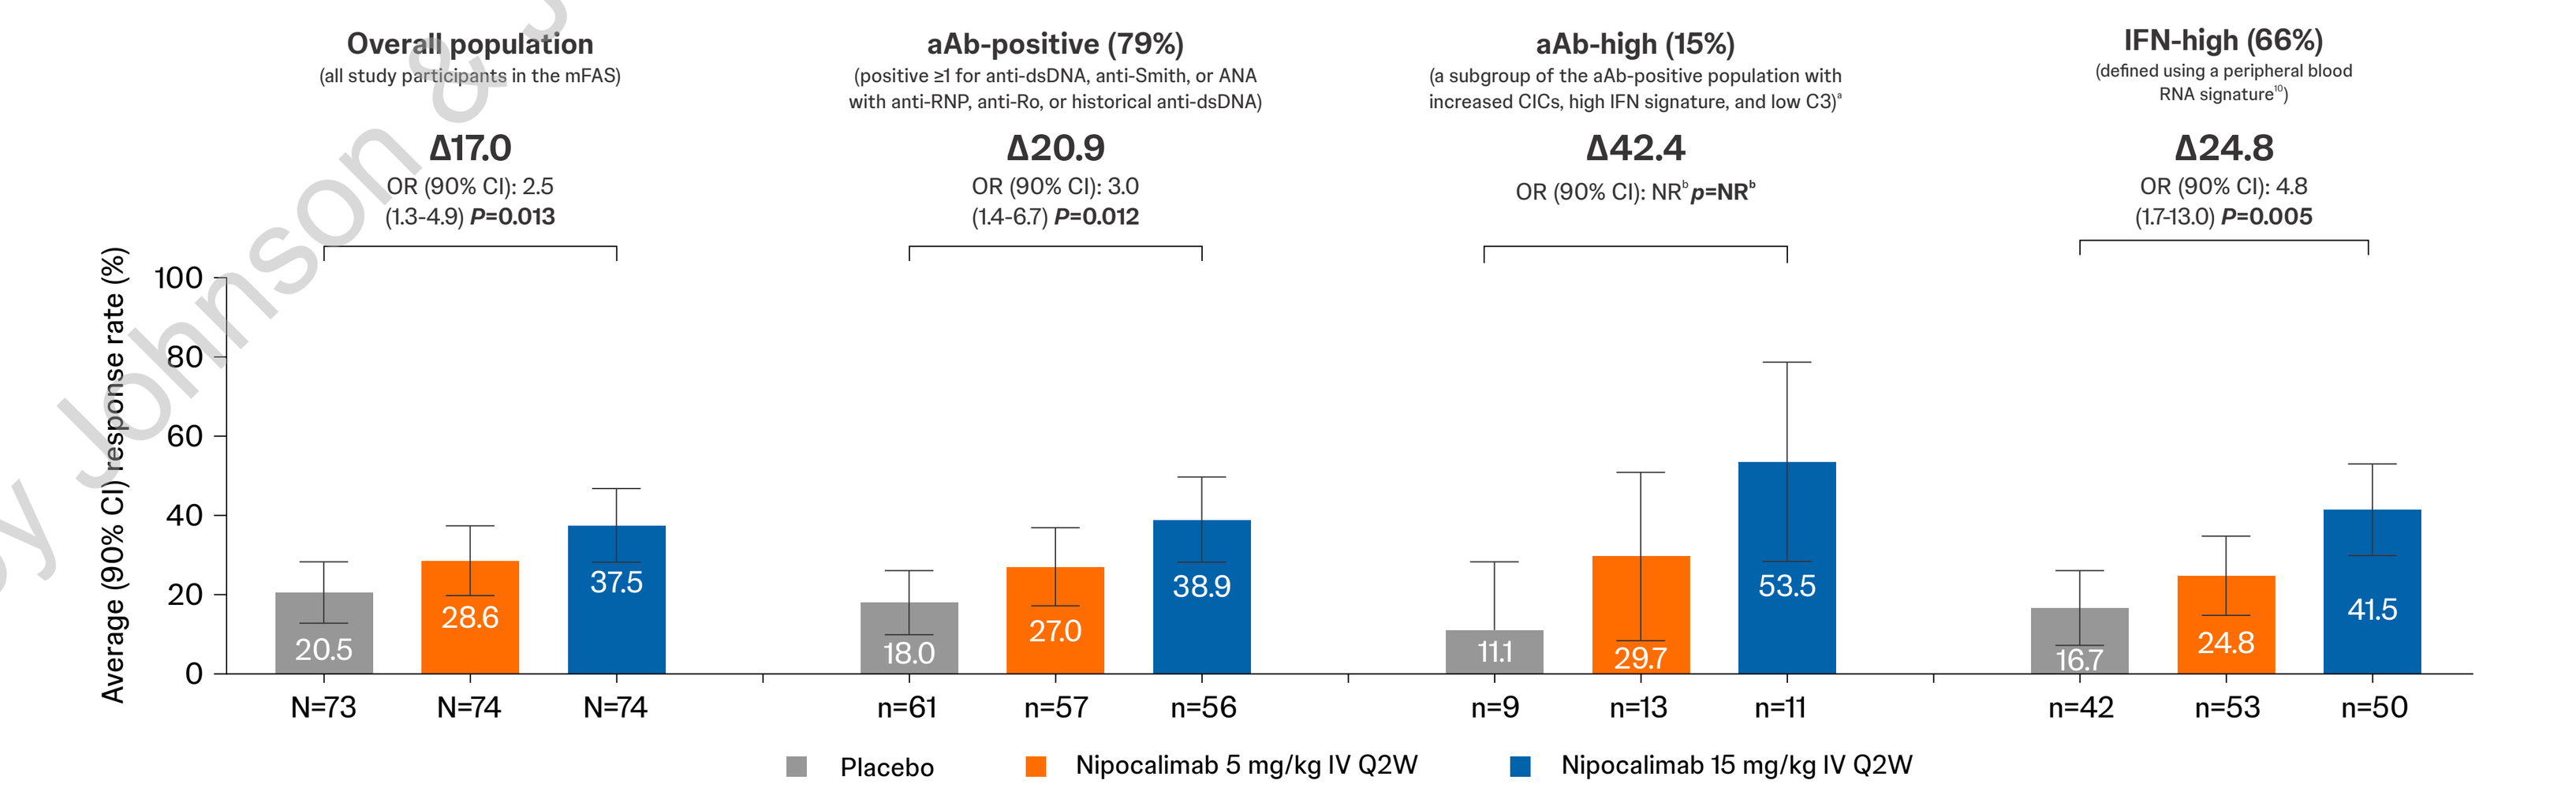
Overall population
(all study participants in the mFAS)
Δ17.0
OR (90% CI): 2.5 (1.3-4.9) P=0.013
aAb-positive (79%)
(positive ≥1 for anti-dsDNA, anti-Smith, or ANA with anti-RNP, anti-Ro, or historical anti-dsDNA)
Δ20.9
OR (90% CI): 3.0 (1.4-6.7) P=0.012
aAb-high (15%)
(a subgroup of the aAb-positive population with increased CICs, high IFN signature, and low C3)[a]
Δ42.4
OR (90% CI): NR[b] p=NR[b]
IFN-high (66%)
(defined using a peripheral blood RNA signature[10])
Δ24.8
OR (90% CI): 4.8 (1.7-13.0) P=0.005
Average (90% CI) response rate (%)
20.5 28.6 37.5
N=73 N=74 N=74
18.0 27.0 38.9
n=61 n=57 n=56
11.1 29.7 53.5
n=9 n=13 n=11
16.7 24.8 41.5
n=42 n=53 n=50
Placebo
Nipocalimab 5 mg/kg IV Q2W
Nipocalimab 15 mg/kg IV Q2W

Analyses based on the mFAS with imputed data. All P values reported here are nominal. [a]Identified through unsupervised consensus clustering based on semiquantitative baseline aAb intensities. [b]Due to small sample sizes and large variability, the OR and P value are NR.

aAb=autoantibody, ANA=antinuclear antibody, C3=complement component 3, CI=confidence interval, CIC=circulating immune complex, dsDNA=double-stranded deoxyribonucleic acid, IFN=interferon, IV=intravenous, LLDAS=Lupus Low Disease Activity State, mFAS=modified full analysis set, NR=not reported, OR=odds ratio, Q2W=every 2 weeks, RNP=ribonucleoprotein.

Nipocalimab reduced total IgG and anti-dsDNA across the treatment period

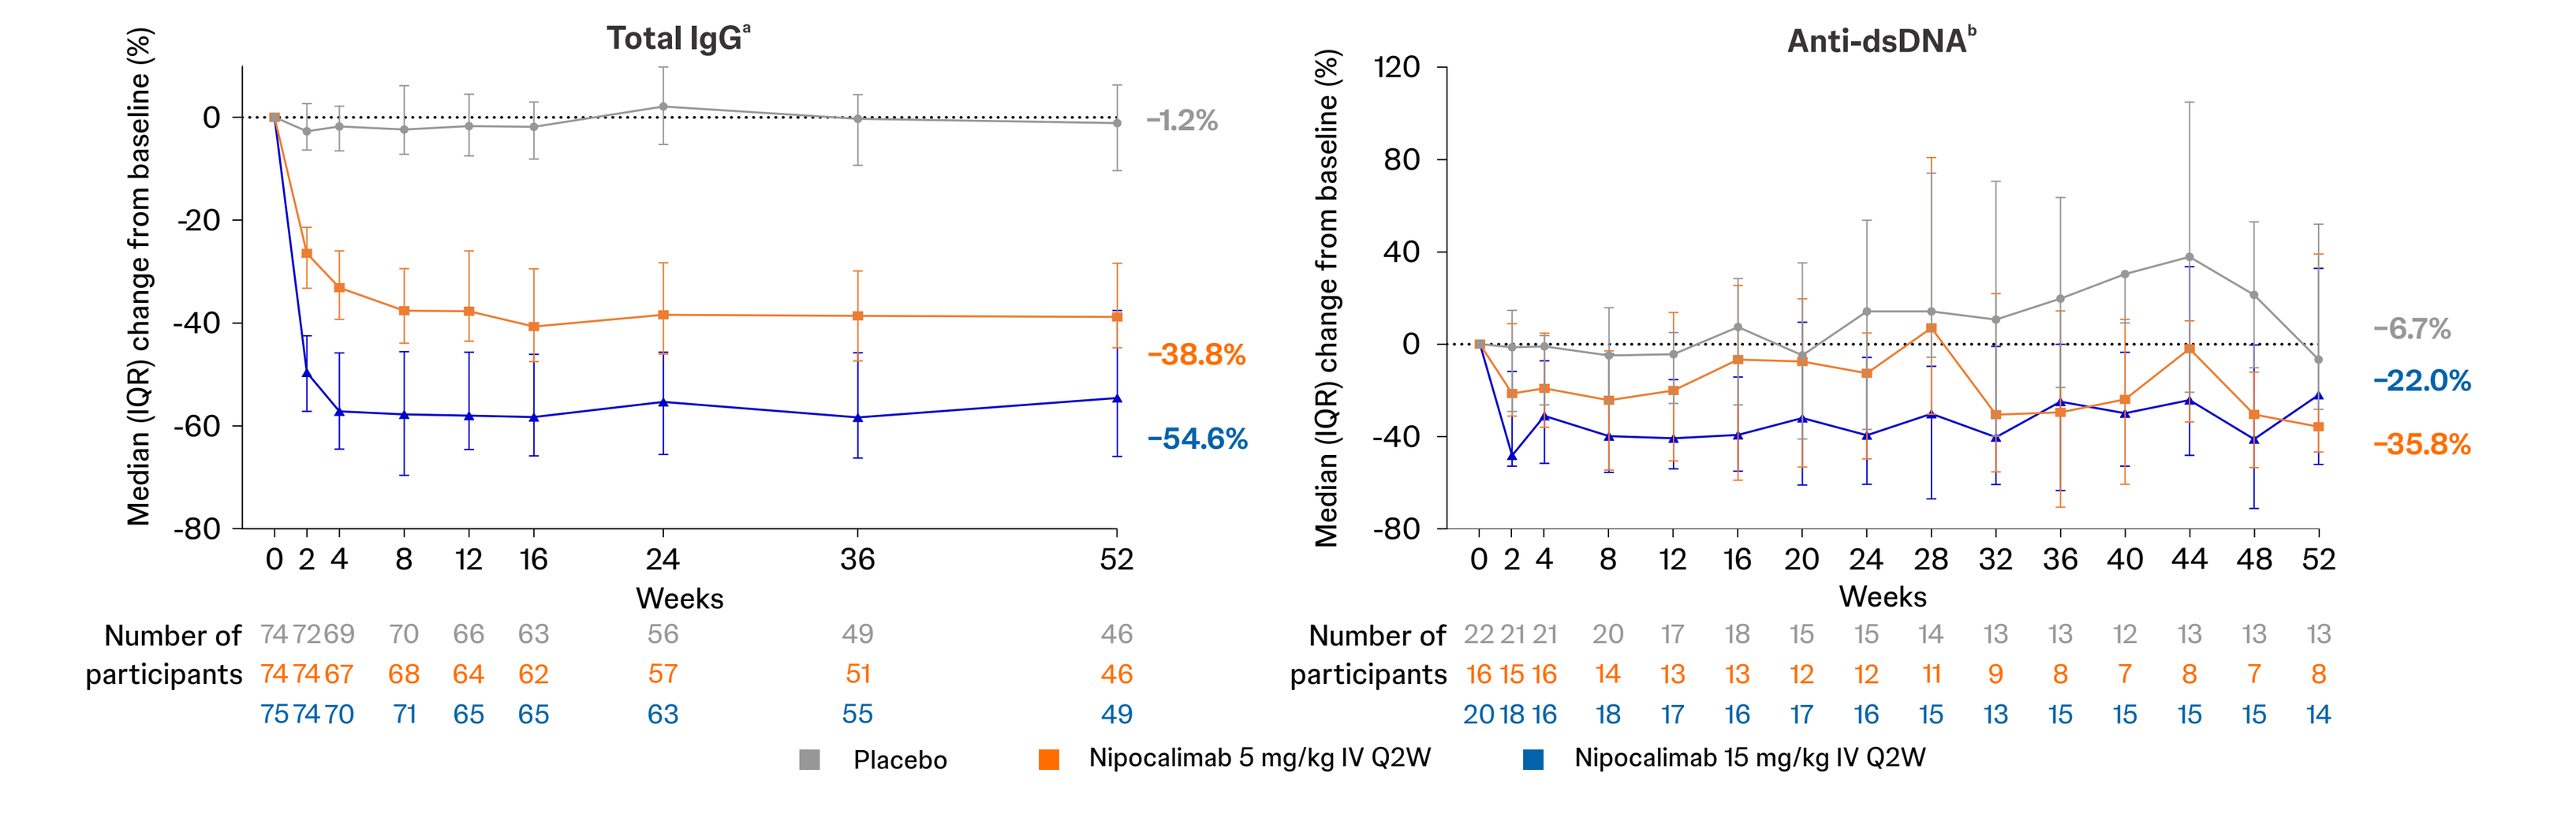
Total IgG[a]
Median (IQR) change from baseline (%)
Weeks
Number of participants
-1.2%
-38.8%
-54.6%
Anti-dsDNA[b]
-6.7%
-22.0%
-35.8%
Placebo
Nipocalimab 5 mg/kg IV Q2W
Nipocalimab 15 mg/kg IV Q2W

Based on the FAS with ≥1 valid postdose blood sample drawn for pharmacodynamic analysis. If a participant missed a planned dose of study intervention at any visit, their data was excluded from all subsequent visits after the first occurrence of a missed dose. [a]20 (27%) participants in the nipocalimab 5 mg/kg group and 47 (63%) in the nipocalimab 15 mg/kg group had minimum IgG <LLN (6 g/L). [b]In anti-dsDNA–positive participants at baseline.

dsDNA=double-stranded deoxyribonucleic acid, FAS=full analysis set, IgG=immunoglobulin G, IQR=interquartile range, IV=intravenous, LLN=lower limit of normal, Q2W=every 2 weeks.

Nipocalimab reduced CIC and increased C3 levels across the treatment period

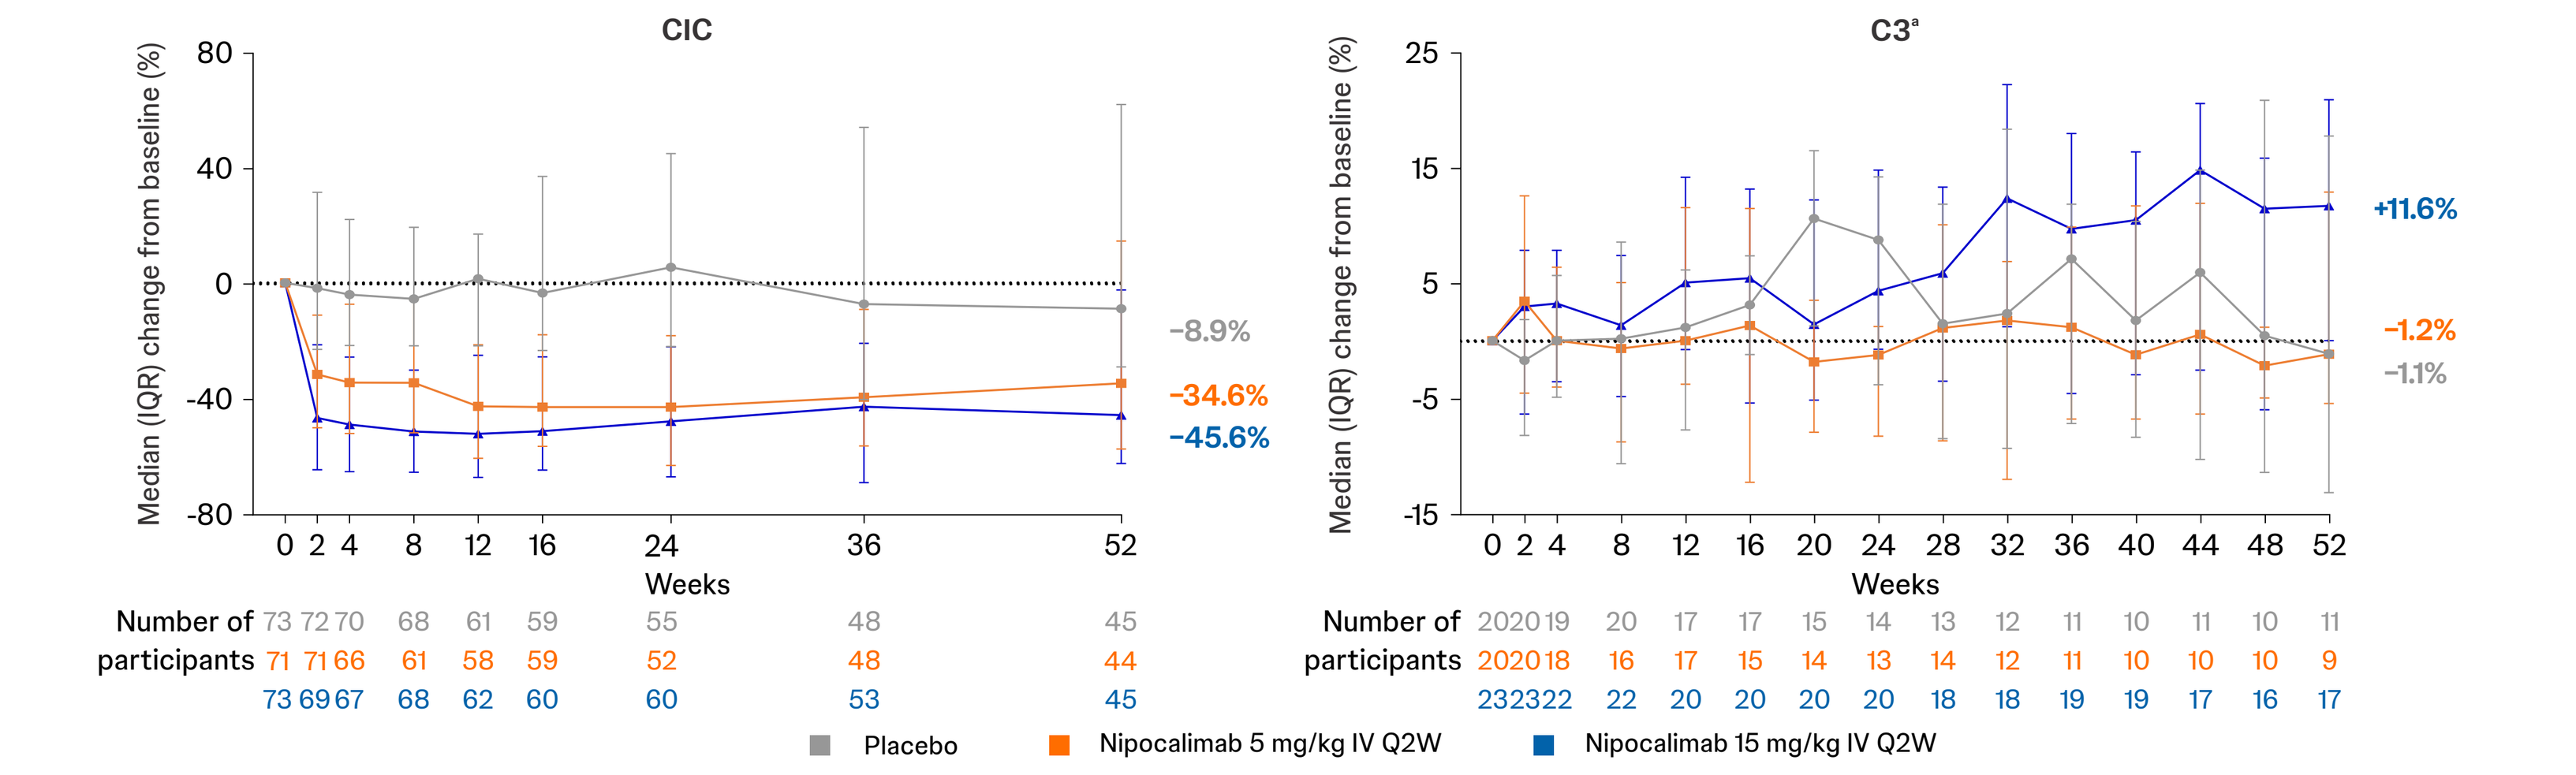
CIC
Median (IQR) change from baseline (%)
Weeks
Number of participants
-8.9%
-34.6%
-45.6%
C3[a]
+11.6%
-1.2%
-1.1%
Placebo
Nipocalimab 5 mg/kg IV Q2W
Nipocalimab 15 mg/kg IV Q2W

Based on the FAS with ≥1 valid postdose blood sample drawn for pharmacodynamic analysis. If a participant missed a planned dose of study intervention at any visit, their data was excluded from all subsequent visits after the first occurrence of a missed dose. [a]In participants with low baseline C3.

C3=complement component 3, CIC=circulating immune complexes, FAS=full analysis set, IgG=immunoglobulin G, IQR=interquartile range, IV=intravenous, Q2W=every 2 weeks.

Nipocalimab was well tolerated, with no new safety signals observed

- Rates of AEs were not dose dependent
- Through Week 58, nadir IgG <3 g/L was observed in 0%, 2.7%, and 22.7% of participants treated with placebo, nipocalimab 5 mg/kg, and nipocalimab 15 mg/kg, respectively. None of the participants with IgG <3 g/L developed serious AEs; 1 participant in the nipocalimab 15 mg/kg IV Q2W group discontinued due to protocol-specified IgG stopping criteria
- No severe hypoalbuminemia (<20 g/L) was observed
- Two deaths were reported in participants from each nipocalimab treatment group due to septic shock. Both events were assessed by the investigator as unrelated to the study intervention

Participants with ≥1 AE,[a] n (%)	Placebo (N=75)	Nipocalimab 5 mg/kg IV Q2W (N=77)	Nipocalimab 15 mg/kg IV Q2W (N=76)	Combined nipocalimab (N=153)
AEs	57 (76.0)	69 (89.6)	63 (82.9)	132 (86.3)
Serious AEs	6 (8.0)	6 (7.8)	10 (13.2)	16 (10.5)
AEs leading to death	0	1 (1.3)	1 (1.3)	2 (1.3)
Infections and infestations	42 (56.0)	45 (58.4)	46 (60.5)	91 (59.5)
Serious infections and infestations	3 (4.0)	4 (5.2)	3 (3.9)	7 (4.6)
Opportunistic infections	0	0	1 (1.3)	1 (0.7)
Infusion reactions	6 (8.0)	10 (13.0)	5 (6.6)	15 (9.8)
Hypersensitivity reactions[b]	9 (12.0)	10 (13.0)	14 (18.4)	24 (15.7)
MACE[c]	1 (1.3)	1 (1.3)	1 (1.3)	2 (1.3)

[a]Based on the FAS. [b]Defined by the Standardized Medical Dictionary for Regulatory Activities (MedDRA) Queries. [c]Cardiovascular death, nonfatal myocardial infarction, and nonfatal stroke.

AE=adverse event, FAS=full analysis set, IgG=immunoglobulin G, IV=intravenous, MACE=major adverse cardiovascular events, Q2W=every 2 weeks.

This material is distributed for scientific purposes by Johnson & Johnson Innovative Medicine and is not for promotional use.

PRESENTED BY: Thanh Nguyen at the Congress of Clinical Rheumatology-West (CCR-West), September 17–20, 2026; Huntington Beach, California, USA.
REFERENCES: 1. Ameer MA, et al. *Cureus.* 2022;14(10):e30330. **2.** Herrada AA, et al. *Front Immunol.* 2019;10:772. **3.** Blumberg LJ, et al. *Sci Adv.* 2019;5(12):eaax9586. **4.** Roopenian DC, Akilesh S. *Nat Rev Immunol.* 2007;7(9):715-725. **5.** Peter HH, et al. *J Allergy Clin Immunol.* 2020;146(3):479-491.e5. **6.** Seth NP, et al. *MAbs.* 2025;17(1):2461191. **7.** Ling LE, et al. *Clin Pharmacol Ther.* 2019;105(4):1031-1039. **8.** Noaiseh G, et al. *Lancet.* 2025;406(10518):2435-2448. **9.** IMAAVY™ (nipocalimab-aahu) injection, for intravenous use [prescribing information]. Janssen Biotech, Inc.; 2025. **10.** Cesaroni M, et al. Type interferon signatures and methods of use. WO2020084591A1. Issued October 25, 2019.
ACKNOWLEDGEMENTS: All study participants, steering committee members, investigators, and study site staff are acknowledged for their contributions to the JASMINE-SLE study. Medical writing support was provided by Sarah Ferrigno, PhD, of Lumanity Communications Inc., and was funded by Johnson & Johnson. Layout design for this poster was provided by Samita Warang (SIRO Medical Writing Pvt. Ltd., India). This study and presentation were sponsored by Johnson & Johnson. **DISCLOSURES: RAF** received personal fees from AstraZeneca, Biogen, Bristol Myers Squibb, GSK, Genentech/Roche, and Johnson & Johnson during the conduct of the study and outside the submitted work; and received grants from Genentech/Roche. **RvV** received consulting fees from AbbVie, AstraZeneca, Biogen, Bristol Myers Squibb, Galapagos, GSK, Instituto Científico Pfizer, Johnson & Johnson, Kyowa Kirin, RemeGen, Sanofi, UCB, and Viela Bio; and received financial support from Bristol Myers Squibb, F. Hoffmann-La Roche, and Merck. **RW** has no conflicts of interest to disclose. **EM** received grant/research support from AbbVie, Amgen, AstraZeneca, Biogen, Bristol Myers Squibb, Eli Lilly, EMD Serono, Genentech, GSK, Johnson & Johnson, and UCB; and received consulting fees from Amgen, AstraZeneca, Biogen, Bristol Myers Squibb, Eli Lilly, EM D Serono, Genentech, GSK, Johnson & Johnson, Servier, UCB, and Wolf Biotherapeutics; and received speaking fees and/or honoraria from AstraZeneca, Eli Lilly, GSK, and Novartis. **GJP-E** received payment or honoraria for advisory board and/or lectures and/or support for attending scientific events from AbbVie, AstraZeneca, Boehringer Ingelheim, GSK, Johnson & Johnson, Novartis, Pfizer, and RemeGen; and received grant/research support from GSK and Johnson & Johnson for conducting the epidemiology study (payment made to GLADEL). **GAK** participated in a speakers bureau and received consulting fees from Bristol Myers Squibb, Johnson & Johnson, and Sanofi -Genzyme-Regeneron; and received grant/research support from Pfizer. **BvH, RWH, TR, SG, RG, KHL, JHL, FZ, SJM, SL, KM, KS, LD, LS, LO, EN, TN** and **FL-W** are employees of Johnson & Johnson and may hold stock or stock options in Johnson & Johnson. **MP** received grant/research support from AstraZeneca, Exagen, and GSK; and received consulting fees from Amgen, Annenberg Center for Health Sciences, AstraZeneca, Blackrock, Eli Lilly, Exagen, Glenmark, GSK, IQVIA, Johnson & Johnson, Merck EMD, Serono, Novartis, Quintiles, Sanofi -Genzyme-Regeneron, Thermo Fisher, and UCB. **PREVIOUSLY PRESNTED AT:** The European Alliance of Associations for Rheumatology (EULAR) European Congress; 3–6 June 2026; London, England, UK.