Nipocalimab for Warm Autoimmune Hemolytic Anemia: Results From the Phase 2/3 Randomized Double-Blind ENERGY Study

Bruno Fattizzo,[1,2] Irina Murakhovskaya,[3] Yasutaka Ueda,[4] Doug Schlichting,[5] Kristen Sweet,[5] Michael Zelasky,[5] Thanh Nguyen,[6] Jagriti Craig,[5] Jocelyn Leu,[5] Leona Ling,[7] Sheryl Pease,[8] Cathye Shu[5]
[1]Hematology Unit, Fondazione IRCCS Ca' Granda Ospedale Maggiore Policlinico, Milan, Italy; [2]Department of Oncology and Hemato-Oncology, University of Milan, Milan, Italy; [3]Albert Einstein College of Medicine/Montefiore Medical Center, Bronx, NY, USA; [4]The University of Osaka, Suita, Osaka, Japan; [5]Johnson & Johnson, Spring House, PA, USA; [6]Johnson & Johnson, Horsham, PA, USA; [7]Johnson & Johnson, Cambridge, MA, USA; [8]Johnson & Johnson, Raritan, NJ, USA

Scan the QR code.
The QR code is intended to provide scientific information for individual reference, and the information should not be altered or reproduced in any way.

This material distributed for scientific purposes by Johnson & Johnson Innovative Medicine and is not for promotional use.

Key Takeaways

- **Nipocalimab, an FcRn blocker that can selectively lower IgG including pathogenic autoantibodies, demonstrated consistent and clinically meaningful benefits across multiple endpoints**
- **Durable hemoglobin response:** Nipocalimab 30 mg/kg q4w showed a significant durable hemoglobin response and favorable responses in prespecified supporting hemoglobin measures
- **Rapid, maintained improvement in hemoglobin:** Hemoglobin increase was rapid, with 1 g/dL improvement observed at Week 1 and maintained over 24 weeks
- **Fatigue improvement:** Nipocalimab 30 mg/kg q4w showed improvements in fatigue were evident as early as Week 2 and maintained through Week 24
- **Consistent, well-tolerated safety:** Nipocalimab was generally well tolerated with no new safety signals identified. Occurrence of grade ≥3 infections were no higher than placebo, and no hypogammaglobulinemia was observed

Background

- Warm autoimmune hemolytic anemia (wAIHA) is a rare, life-threatening, relapsing-remitting disease characterized by autoantibody-mediated premature destruction of red blood cells (RBCs)[1–3]
- Debilitating fatigue is the primary patient-reported symptom of wAIHA, and is identified by patients as the most debilitating symptom, limiting daily functioning and often requiring frequent rest[4,5]
 - Main complications include infections and thrombosis[2]
 - 5-year mortality is ~30%[6]
- No treatments are currently approved for wAIHA; current management involves off-label, broadly immunosuppressive regimens associated with relapse and adverse events (AEs)
 - Corticosteroids: two-thirds of patients relapse or do not respond[2,3]
 - Rituximab: half of patients relapse; 2 to 6 months to induce a response[7,8]
- Nipocalimab is an immunoselective neonatal Fc receptor (FcRn) blocker that reduces circulating immunoglobulin G (IgG) levels, including pathogenic autoantibodies, while preserving immune function[9–13]
 - In patients with wAIHA, nipocalimab lowers anti-RBC pathogenic antibodies and has the potential of decreasing hemolysis and increasing hemoglobin[14]

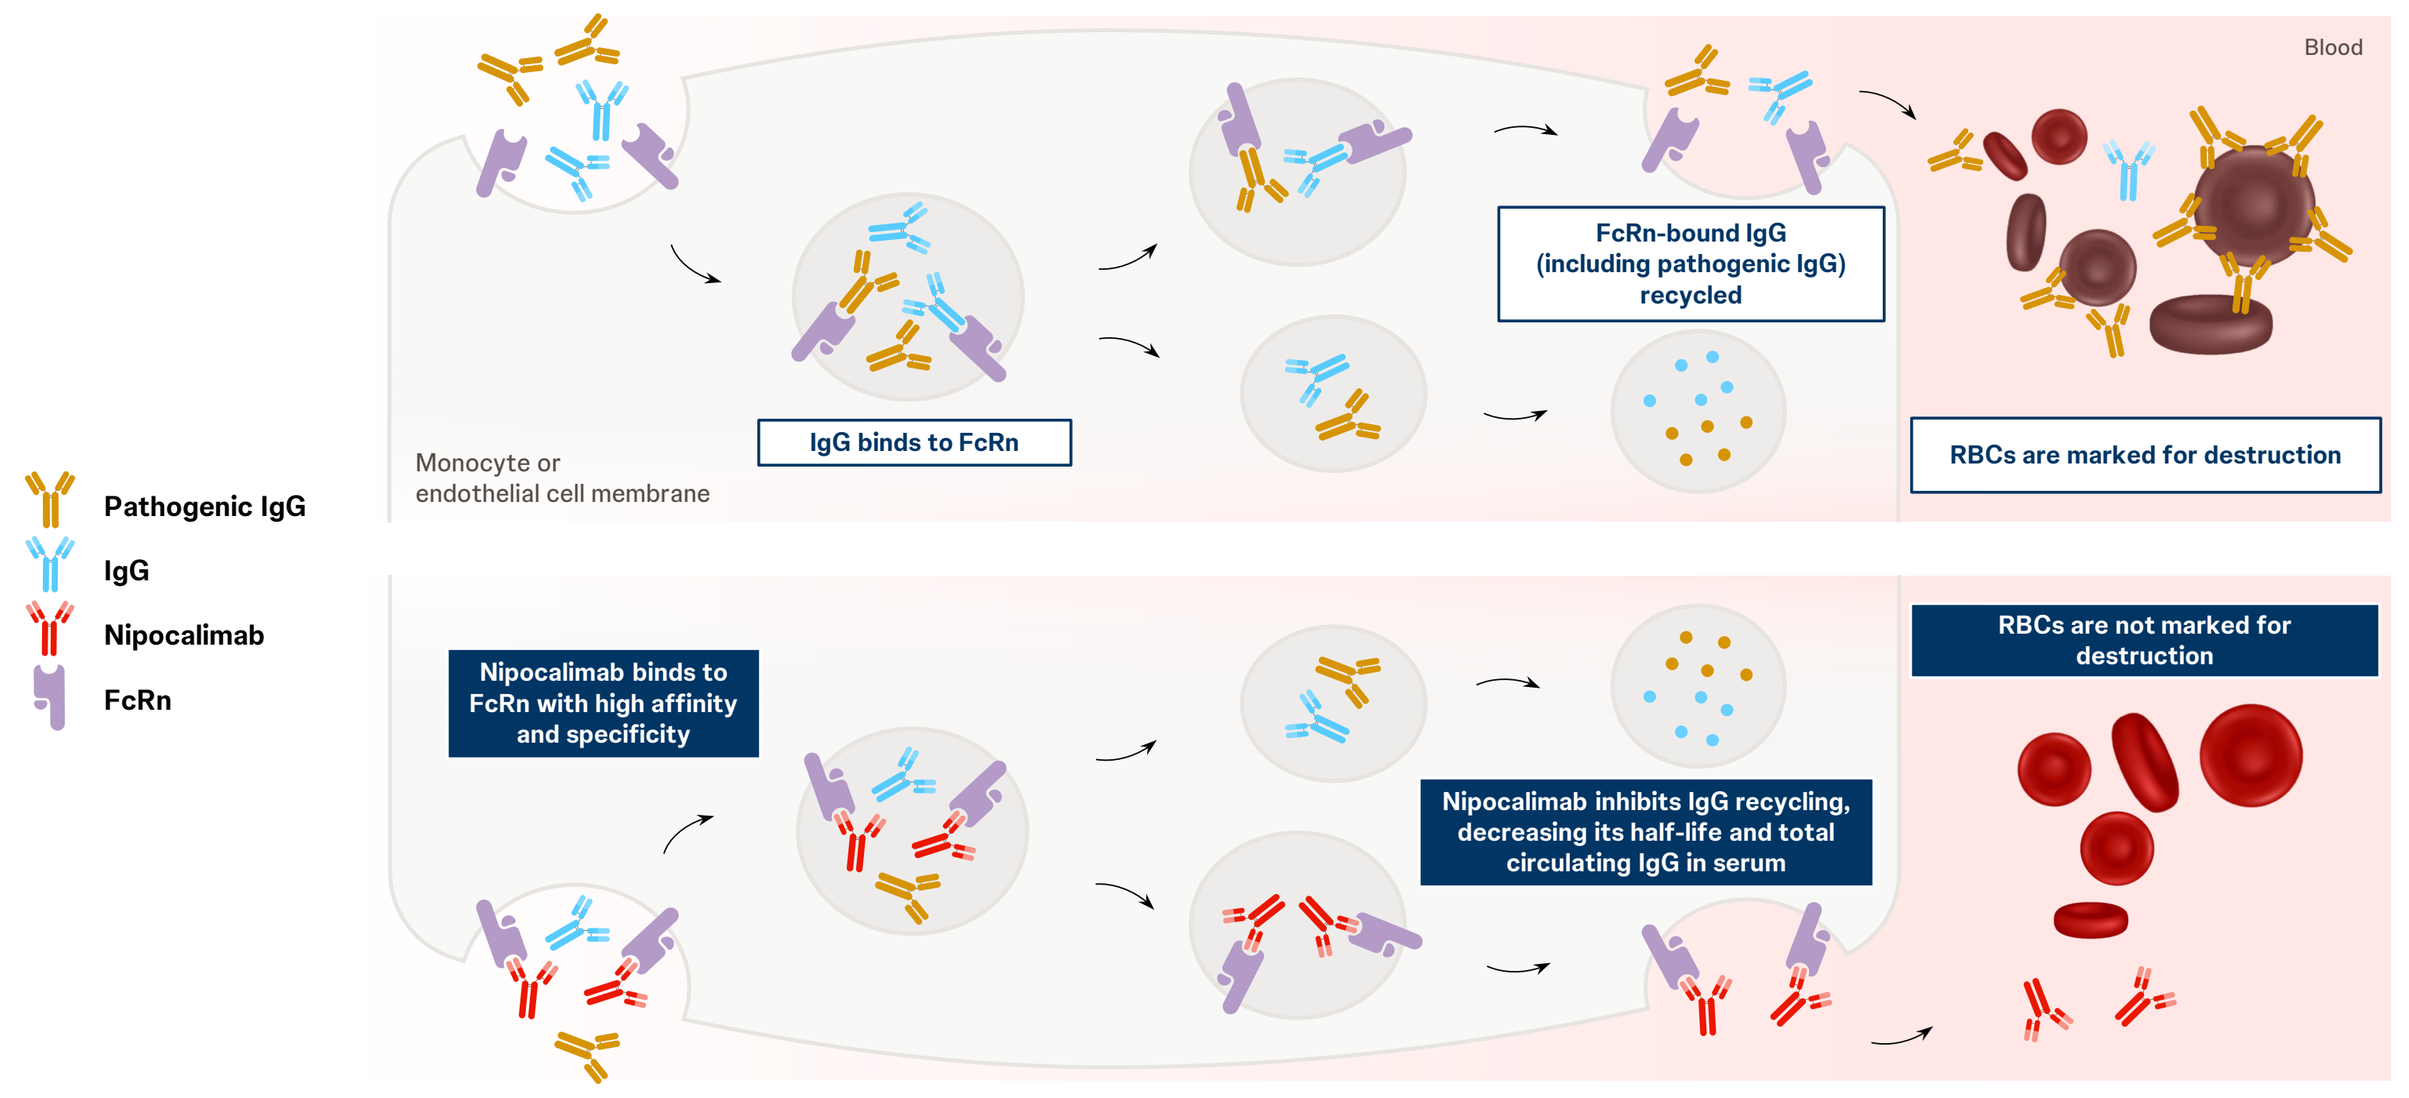
Pathogenic IgG
IgG
Nipocalimab
FcRn
Blood
Monocyte or endothelial cell membrane
IgG binds to FcRn
FcRn-bound IgG (including pathogenic IgG) recycled
RBCs are marked for destruction
Nipocalimab binds to FcRn with high affinity and specificity
Nipocalimab inhibits IgG recycling, decreasing its half-life and total circulating IgG in serum
RBCs are not marked for destruction

FcRn=neonatal Fc receptor, IgG=immunoglobulin G, RBC=red blood cell.

Objective

To evaluate the safety and efficacy of nipocalimab versus placebo for the treatment of wAIHA

Methods

Participants

- ≥18 years of age
- Primary or secondary wAIHA (for ≥3 months)
- Current or previous treatment
- Hgb <10 g/dL
- Positive DAT (IgG or IgG+C3d)
- Evidence of hemolysis

Background treatment

- Stable doses of corticosteroids and/or immunosuppressants allowed
- Those on corticosteroids meeting the primary endpoint are required to taper corticosteroids by 10% q2w

Study Design

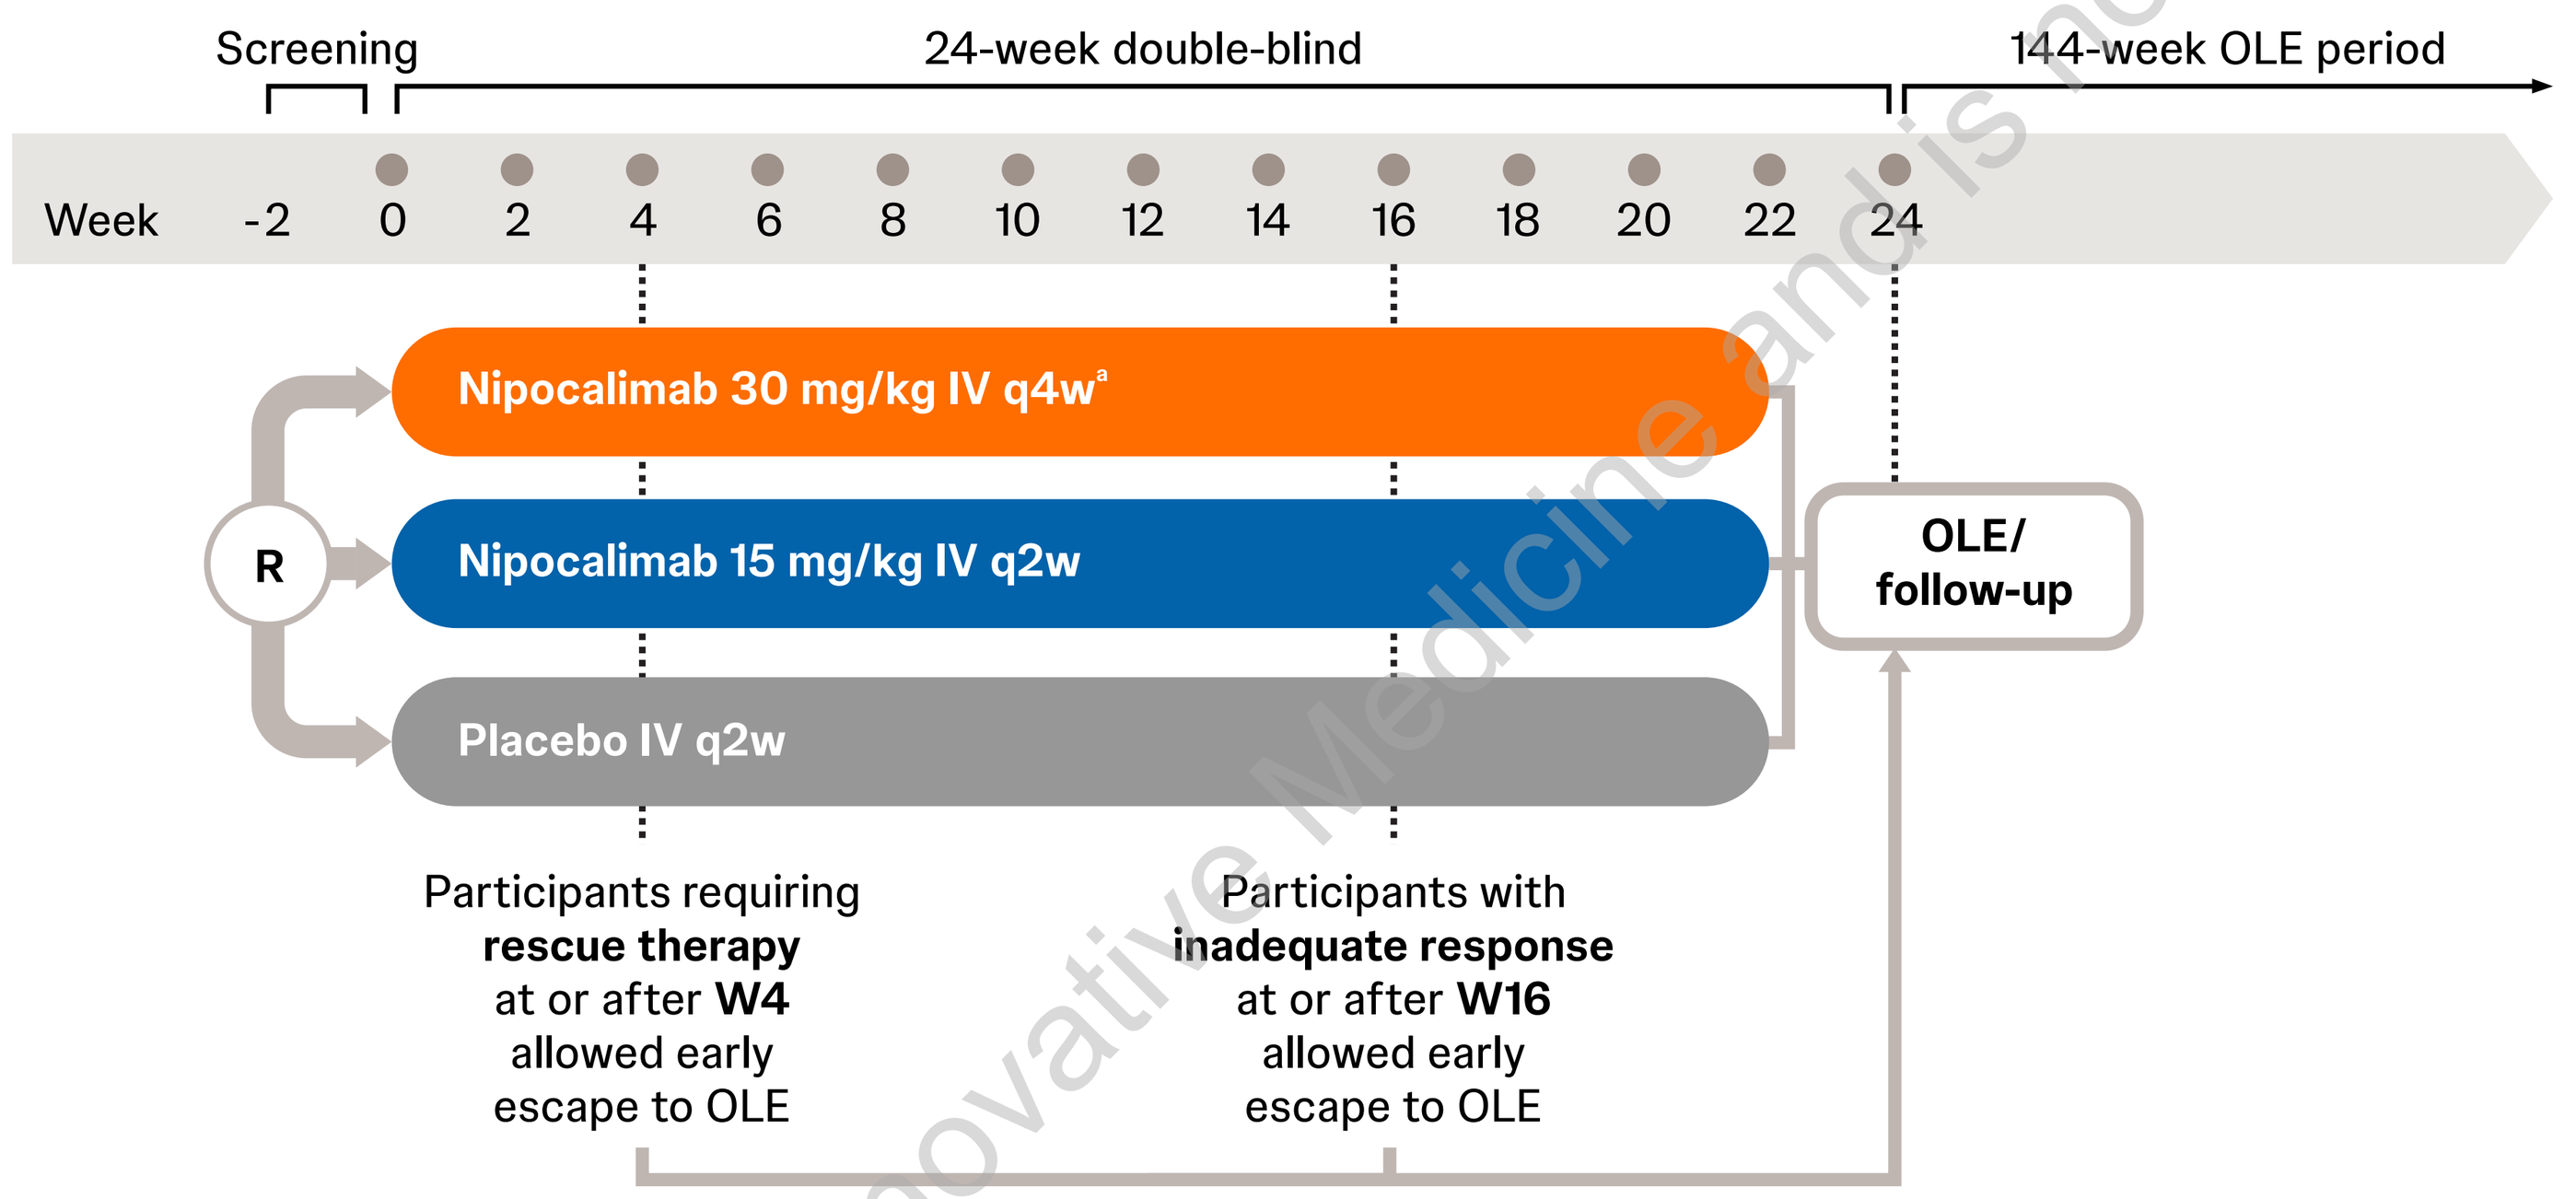
Screening
24-week double-blind
144-week OLE period
Week -2 0 2 4 6 8 10 12 14 16 18 20 22 24
R
Nipocalimab 30 mg/kg IV q4w[a]
Nipocalimab 15 mg/kg IV q2w
Placebo IV q2w
OLE/ follow-up
Participants requiring rescue therapy at or after W4 allowed early escape to OLE
Participants with inadequate response at or after W16 allowed early escape to OLE

ClinicalTrials.gov identifier: NCT04119050. [a]Nipocalimab 30 mg/kg IV at Week 0 and q4w through Week 20 and placebo IV at Week 2 and q4w through Week 22. C3d=complement component 3d, DAT=direct antigen test, Hgb=hemoglobin, IgG=immunoglobulin G, IV=intravenous, OLE=open-label extension, q2w=every 2 weeks, q4w=every 4 weeks, R=randomized, wAIHA=warm autoimmune hemolytic anemia, W=week.

End point assessment

Primary Endpoint: Durable Hemoglobin Response

- Hgb ≥10 g/dL, **AND** Hgb increase ≥2 g/dL from baseline, **AND** for 3 consecutive visits (≥28 days), **AND** starting by Week 16, **AND**
- Without rescue therapy or dose modification of ongoing stable background treatment for wAIHA, including any increase in oral corticosteroids

Key Secondary Endpoints

- Improvement in FACIT-Fatigue (Week 24)
- Corticosteroid dose reduction (Week 24; % reduction in daily corticosteroid dose[b])

[b]Prednisone equivalent.
FACIT-Fatigue=Functional Assessment of Chronic Illness Therapy – Fatigue, Hgb=hemoglobin, wAIHA=warm autoimmune hemolytic anemia.

Results

Baseline characteristics were consistent with the wAIHA patient population

Baseline characteristics	Nipocalimab 30 mg/kg q4w (n=38)	Nipocalimab 15 mg/kg q2w (n=38)	Placebo (n=39)
Mean age (range), years	56.1 (24–79)	53.1 (18–80)	59.9 (24–86)
Female	55%	58%	51%
Primary wAIHA[a]	92%	87%	82%
Median (IQR) time since diagnosis, months	32 (20–60)	44 (21–81)	23 (17–71)
Mean (SD) baseline hemoglobin, g/dL	9.2 (1.4)	8.6 (1.3)	9.0 (1.4)
Mean (SD) FACIT-Fatigue score	34.5 (10.7)	35.0 (10.5)	31.1 (12.2)
Current treatments for wAIHA (%)			
Any concomitant CS	74%	87%	77%
IST or CS at >20 mg/day of prednisone or equivalent[c]	45%	39%	46%
Prior treatments for wAIHA (%)			
Prior rituximab use	42%	55%	49%
Prior transfusions	58%	58%	59%
Prior splenectomy	3%	16%	10%

[a]Stratification factors (primary or versus secondary wAIHA; screening hemoglobin ≤8.5 g/dL or versus >8.5 g/dL; no treatment or corticosteroids at dose ≤20 mg/day of prednisone or equivalent with no immunosuppressants versus immunosuppressants or corticosteroids at >20 mg/day of prednisone or equivalent or no such treatment). CS=corticosteroids, FACIT-Fatigue=Functional Assessment of Chronic Illness Therapy-Fatigue, IQR=interquartile range, IST=immunosuppressive therapy, q2w=every 2 weeks, q4w=every 4 weeks, SD=standard deviation, wAIHA=warm autoimmune hemolytic anemia.

Nipocalimab 30 mg/kg q4w significantly improved durable hemoglobin response versus placebo (Primary endpoint)

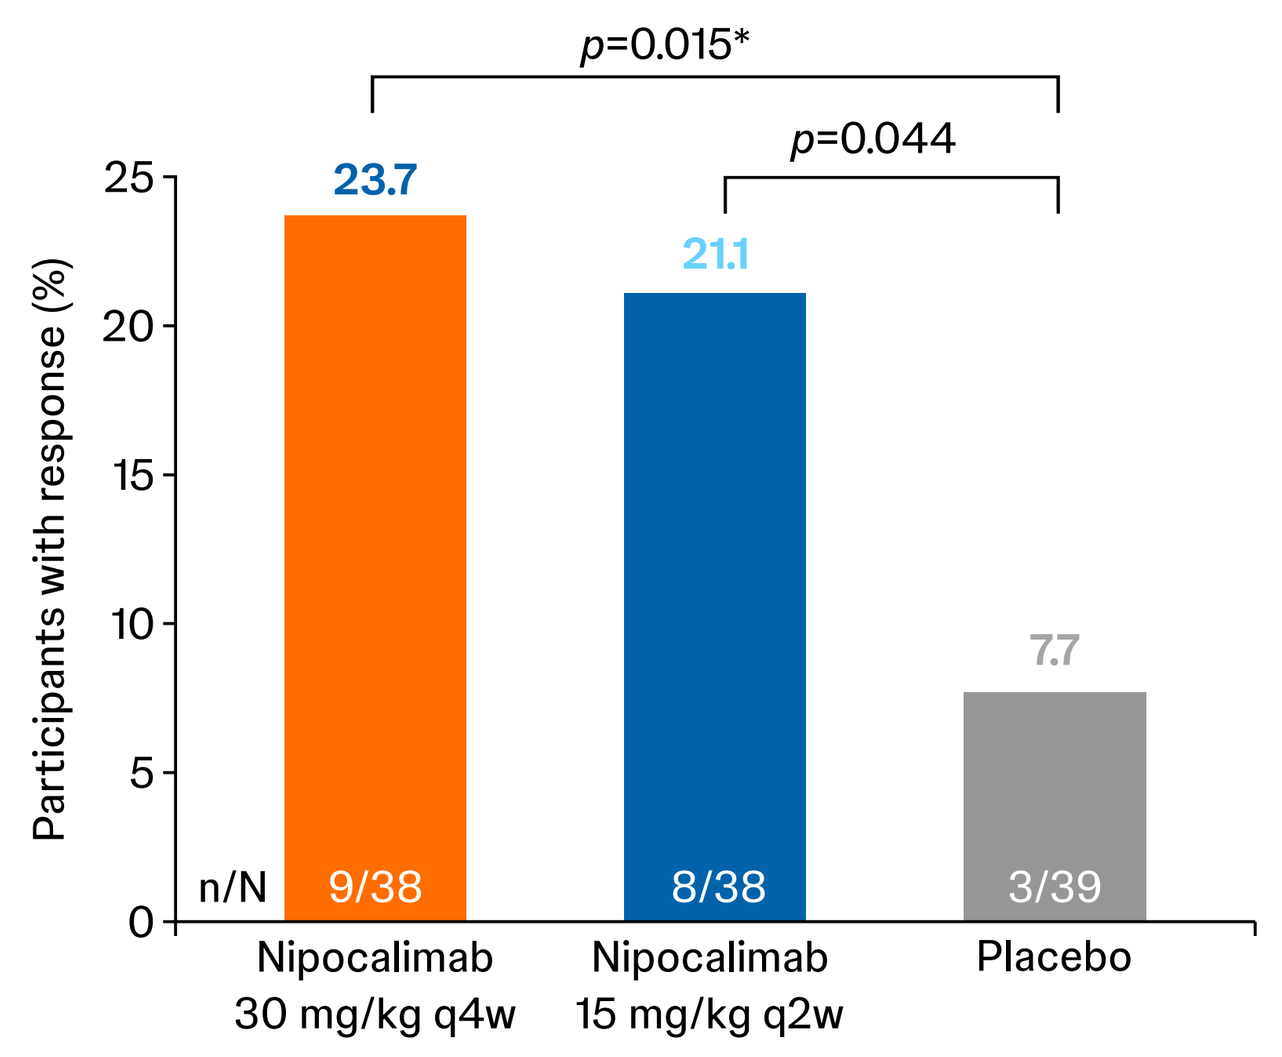
p=0.015*
p=0.044
Participants with response (%)
23.7
21.1
7.7
n/N 9/38
8/38
3/39
Nipocalimab 30 mg/kg q4w
Nipocalimab 15 mg/kg q2w
Placebo

*Durable hemoglobin response defined as: Hgb ≥10 g/dL, AND Hgb increase ≥2 g/dL from baseline, AND for 3 consecutive visits (≥28 days), AND starting by Week 16, AND Without rescue therapy or dose modification of ongoing stable background treatment for wAIHA, including any increase in oral corticosteroids,. Composite analysis strategy (participants with intercurrent events [including any use of rescue therapies or initiation or dose increase of protocol-specified standard-of-care background therapy] were considered to be nonresponders). P values based on equal-weight approach to control the family-wise type I error rate. *One-sided p<0.02499 versus placebo considered significant. Hgb=hemoglobin, q2w=every 2 weeks, q4w=every 4 weeks, wAIHA=warm autoimmune hemolytic anemia.*

Hemoglobin improved by week 1 and was maintained through week 24 in nipocalimab treatment groups

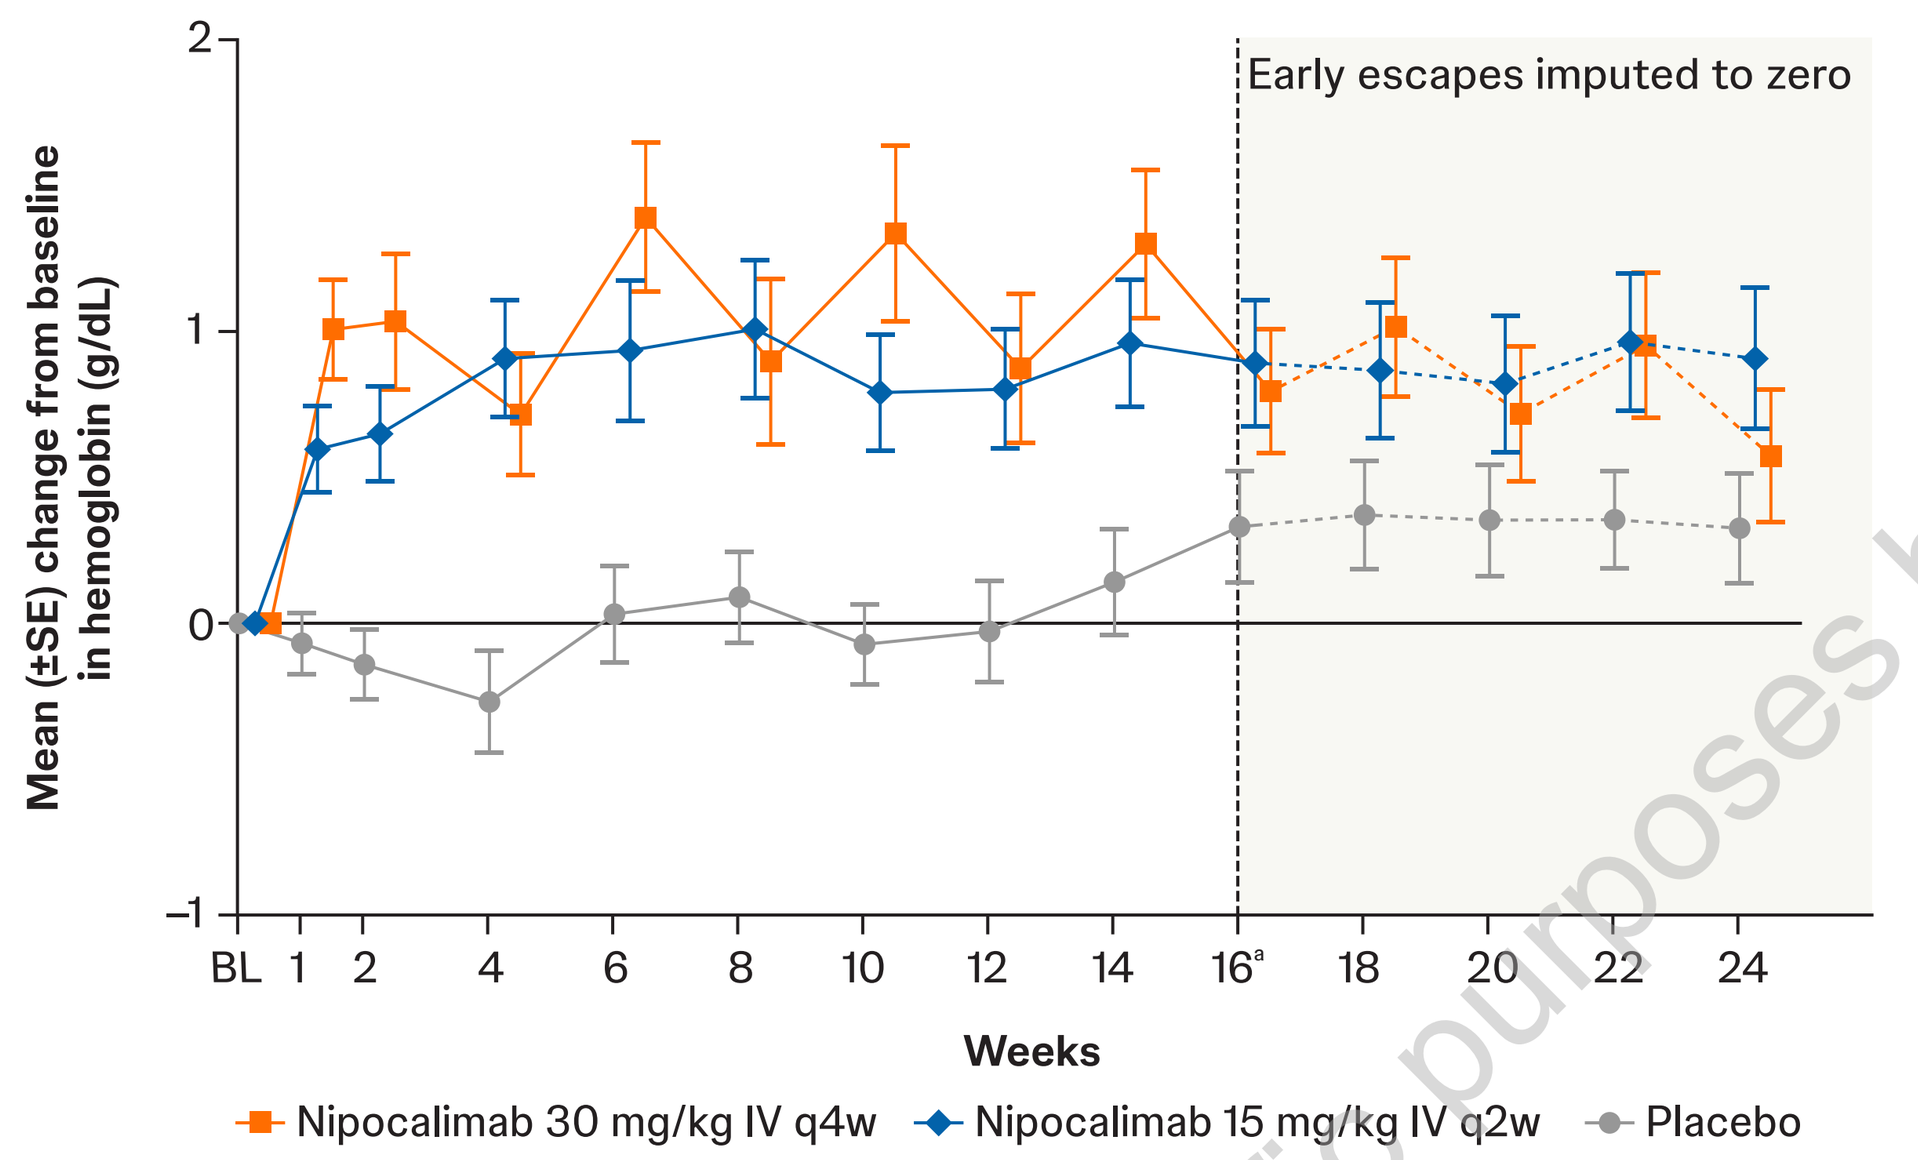
Mean (±SE) change from baseline in hemoglobin (g/dL)
Early escapes imputed to zero
BL 1 2 4 6 8 10 12 14 16[a] 18 20 22 24
Weeks
Nipocalimab 30 mg/kg IV q4w
Nipocalimab 15 mg/kg IV q2w
Placebo

[a]Participants meeting failure criteria (ie, presence of symptoms and failure to demonstrate ≥1 g/dL hemoglobin increase) at or after Week 16 were, at the investigator's discretion, allowed discontinuation of double-blinded treatment and early escape to the OLE. IV=intravenous, q2w=every 2 weeks, q4w=every 4 weeks, SE=standard error.

Prespecified primary endpoint analyses support durable hemoglobin response

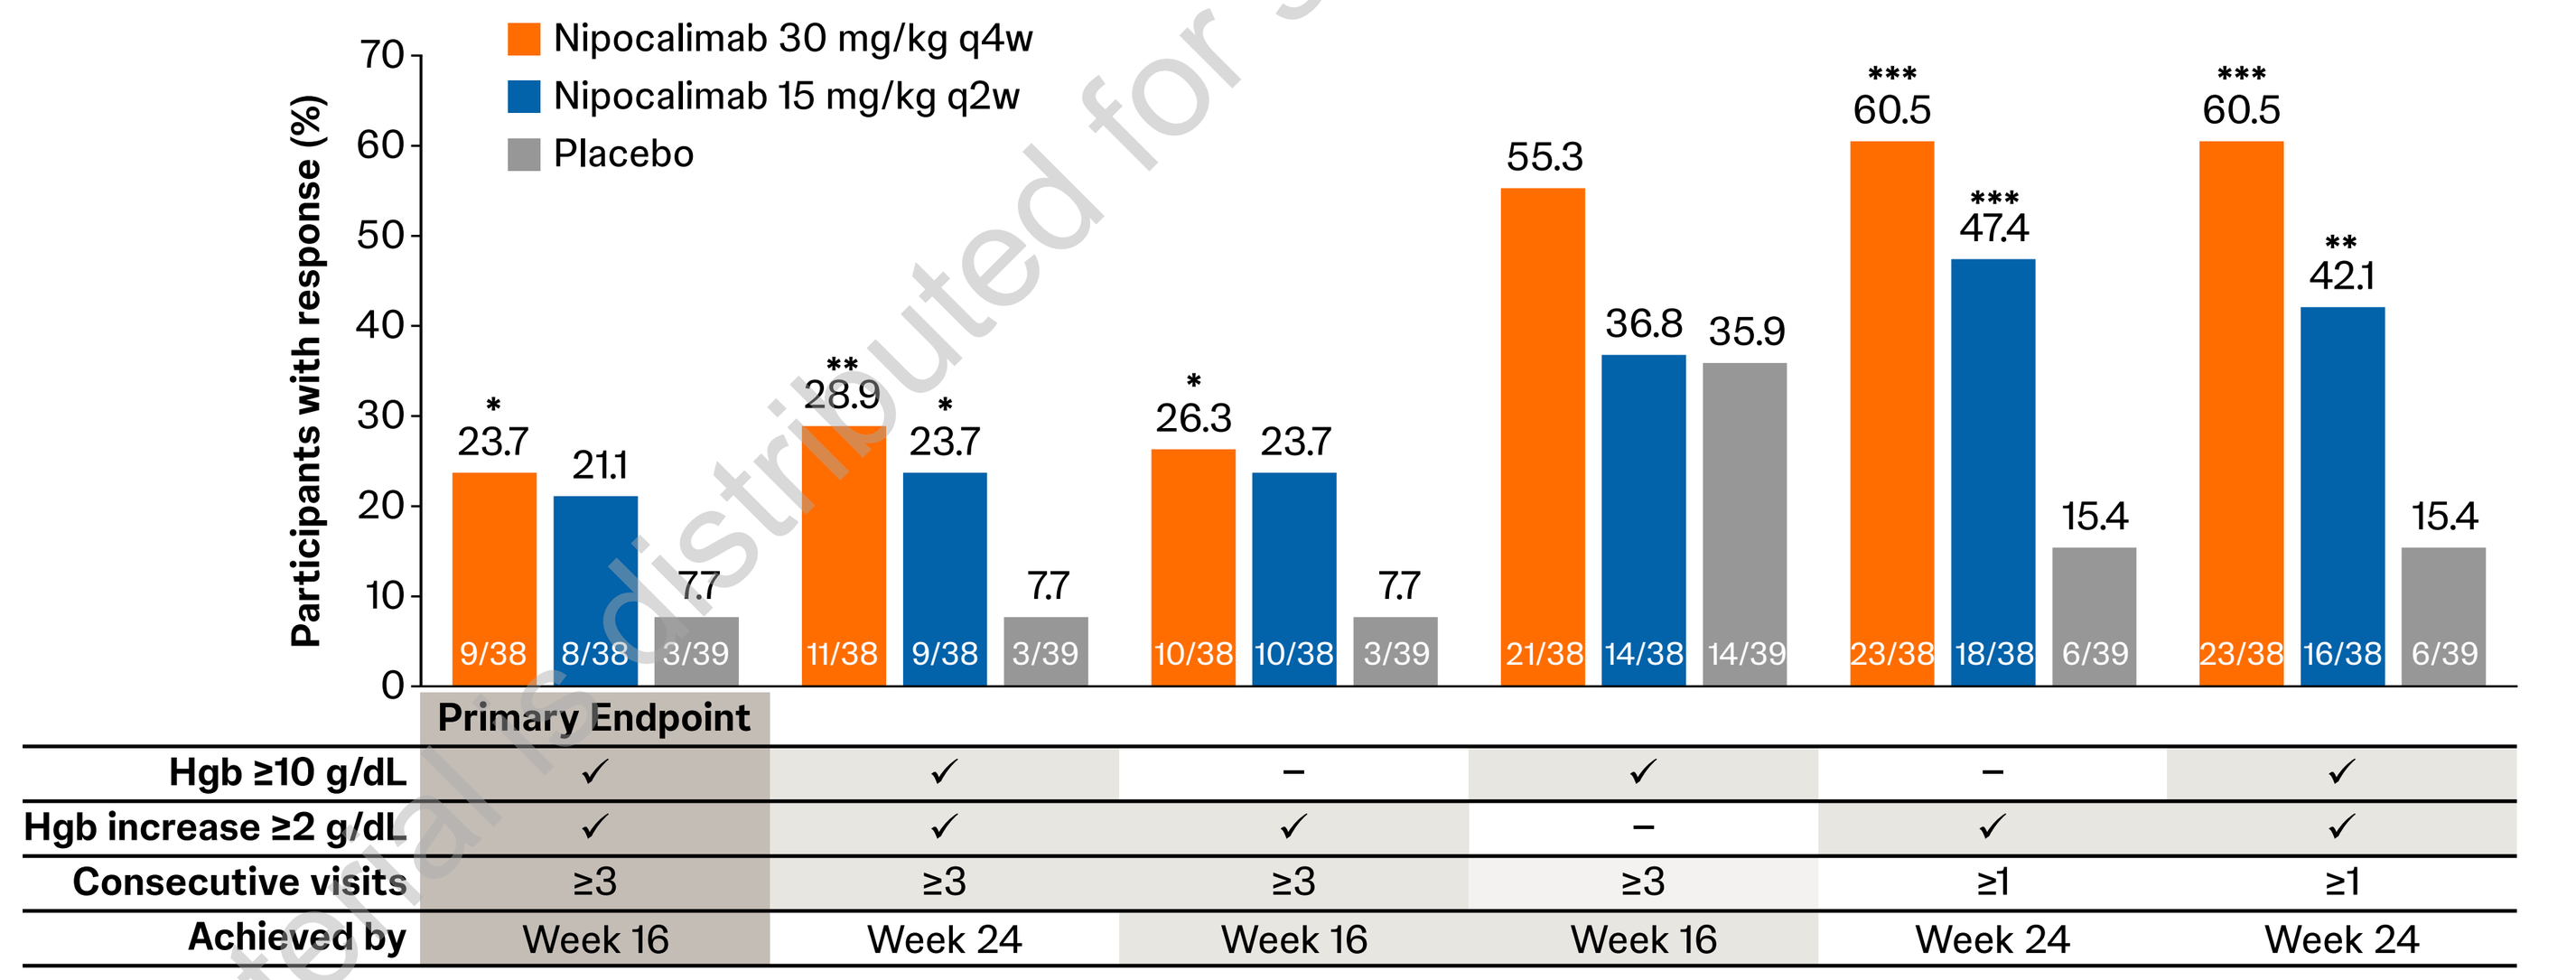
Nipocalimab 30 mg/kg q4w
Nipocalimab 15 mg/kg q2w
Placebo
Participants with response (%)
23.7 21.1 7.7
28.9 23.7 7.7
26.3 23.7 7.7
55.3 36.8 35.9
60.5 47.4 15.4
60.5 42.1 15.4
9/38 8/38 3/39
11/38 9/38 3/39
10/38 10/38 3/39
21/38 14/38 14/39
23/38 18/38 6/39
23/38 16/38 6/39
Primary Endpoint
Hgb ≥10 g/dL ✓ ✓ – ✓ – ✓
Hgb increase ≥2 g/dL ✓ ✓ ✓ – ✓ ✓
Consecutive visits ≥3 ≥3 ≥3 ≥3 ≥1 ≥1
Achieved by Week 16 Week 24 Week 16 Week 16 Week 24 Week 24

*Except for the primary endpoint, all p values are nominal. Composite analysis strategy (participants with intercurrent events [including any use of rescue therapies or initiation or dose increase of protocol-specified standard-of-care background therapy] were considered to be nonresponders). *p<0.02499, **p<0.01, ***p<0.001 versus placebo. Hgb=hemoglobin, q2w=every 2 weeks, q4w=every 4 weeks.*

Fatigue reduction from baseline to week 24 was nominally significant for nipocalimab 30 mg/kg q4w (p=0.007) versus placebo

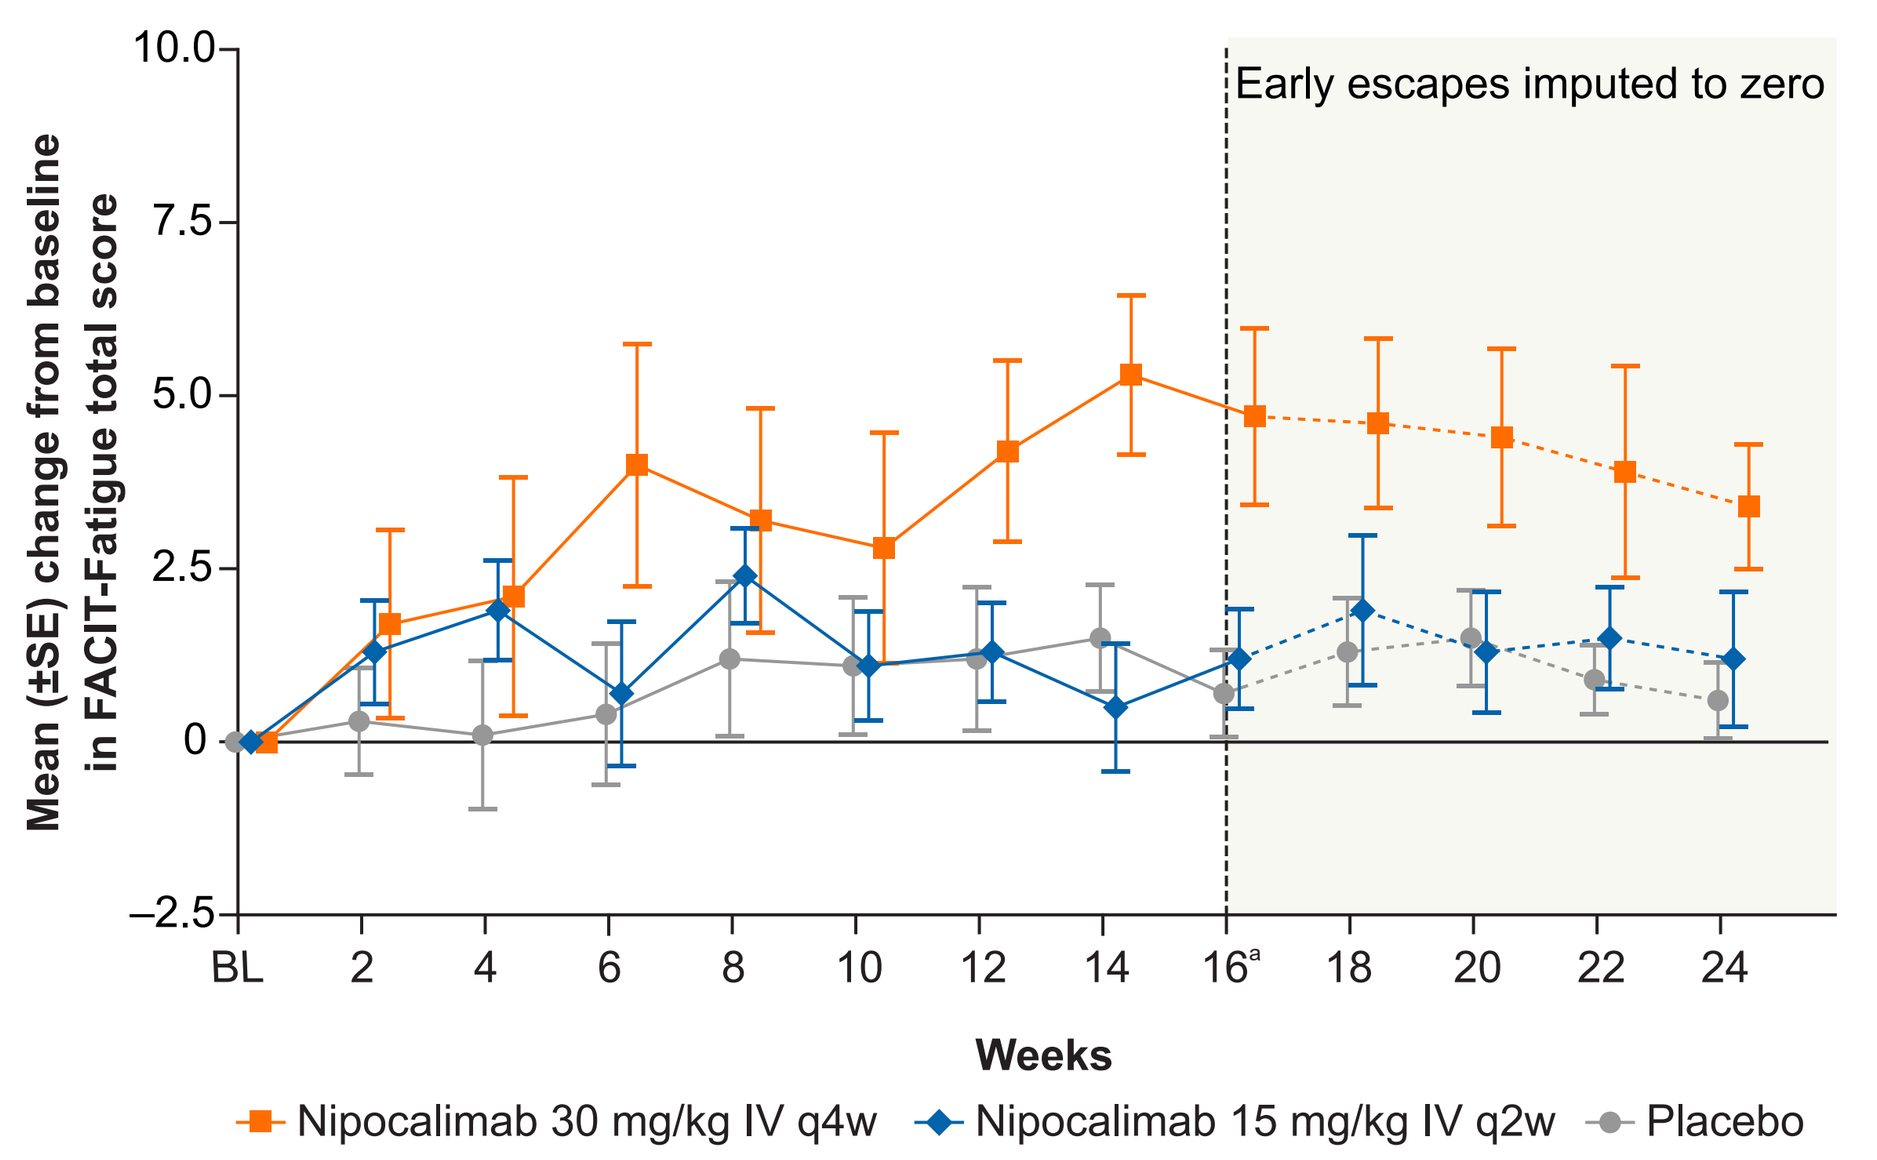
Mean (±SE) change from baseline in FACIT-Fatigue total score
Early escapes imputed to zero
BL 2 4 6 8 10 12 14 16[a] 18 20 22 24
Weeks
Nipocalimab 30 mg/kg IV q4w
Nipocalimab 15 mg/kg IV q2w
Placebo

[a]Participants meeting failure criteria (ie, presence of symptoms and failure to demonstrate ≥1 g/dL hemoglobin increase) at or after Week 16 were, at the investigator's discretion, allowed discontinuation of double-blinded treatment and early escape to the OLE. BL=baseline, FACIT-Fatigue=Functional Assessment of Chronic Illness Therapy – Fatigue, OLE=open-label extension, q2w=every 2 weeks, q4w=every 4 weeks, SE=standard error.

More participants experienced corticosteroid[a] reductions at week 24 with nipocalimab versus placebo

	Nipocalimab 30 mg/kg q4w (n=38)	Nipocalimab 15 mg/kg q2w (n=38)	Placebo (n=39)
Patients taking corticosteroids at baseline, n	26	32	28
Mean change from baseline, mg (SD)	-3.7 (8.3)	-2.5 (6.8)	-0.7 (2.7)
Mean change from baseline, % (SD)	-15.1 (28.24)	-14.0 (30.42)	-3.9 (16.33)
One-sided p value (based on LS means on ranks)	0.039	0.055	-

All p values are nominal, based on an equal-weight approach to control the family-wise type I error rate. Composite analysis strategy (participants with intercurrent events [including any use of rescue therapies or initiation or dose increase of protocol-specified standard-of-care background therapy] were considered to be nonresponders). [a]Participants meeting failure criteria (ie, presence of symptoms and failure to demonstrate ≥1 g/dL hemoglobin increase) at or after Week 16 were, at the investigator's discretion, allowed discontinuation of double-blinded treatment and early escape to the OLE. LS=least squares, OLE=open-label extension, q2w=every 2 weeks, q4w=every 4 weeks, SD=standard deviation.

Safety was consistent with the known safety profile of nipocalimab and the known clinical risks associated with wAIHA

- The majority of treatment emergent adverse events (TEAEs) were grade 2 or 3
- The most common AEs (≥10%) among all participants in the combined nipocalimab group were wAIHA (37.2%), diarrhea (11.5%), and fatigue and pyrexia (both 10.3%); the most common AEs among participants in the placebo group were wAIHA (51.3%) and COVID-19 and nasopharyngitis (both 10.3%)
- No participant experienced hypogammaglobulinemia
- By preferred term, no TEAE leading to discontinuation was reported in more than 1 participant
- Two cases of major adverse cardiovascular event (MACE) occurred in participants with complex medical histories in the nipocalimab 15 mg/kg q2w group, later leading to death (causes of death were not related to study treatment)

AE, n (%)	Nipocalimab 30 mg/kg q4w (n=38)	Nipocalimab 15 mg/kg q2w (n=37)[a]	Placebo (n=39)
TEAEs	35 (92.1)	30 (81.1)	35 (89.7)
SAEs	8 (21.1)	6 (16.2)	14 (35.9)
AEs leading to discontinuation	2 (5.3)	5 (13.5)	1 (2.6)
AEs of interest			
Grade ≥3 infections	2 (5.3)	3 (8.1)	5 (12.8)
Adjudicated MACE	0	2 (5.4)	0
Adjudicated VTE (DVT and/or PE)	1 (2.6)	0	0
Hypoalbuminemia (albumin <20 g/L)	0	0	0
Death	0	2 (5.4)	0

[a]One patient was randomized to nipocalimab 15 mg/kg q2w, but did not receive nipocalimab. AE=adverse event, DVT=deep vein thrombosis, IgG=immunoglobulin G, MACE=major adverse cardiovascular event, PE=pulmonary embolism, q2w=every 2 weeks, q4w=every 4 weeks, SAE=serious adverse event, TEAE=treatment emergent adverse event, wAIHA=warm autoimmune hemolytic anemia, VTE=venous thromboembolism.

PRESENTED BY: Thanh Nguyen at the Congress of Clinical Rheumatology-West (CCR-West), September 17–20, 2026; Huntington Beach, California, USA. **REFERENCES: 1.** Barcellini W. *Transfus Med Hemother.* 2015;42(5):287–293. **2.** Brodsky RA. *N Engl J Med.* 2019;381(7):647–654. **3.** Kuter DJ. *Hematology Am Soc Hematol Educ Program.* 2022;2022(1):105–113. **4.** Shukla A, et al. *Orphanet J Rare Dis.* 2025;20(1):403. **5.** Pease S, et al. *J Patient Rep Outcomes.* 2025;9(1):97. **6.** Cherif H, et al. *Eur J Haematol.* 2025;114(1):129–138. **7.** Murakhovskaya I. *Clin Med.* 2020;9(12):4034. **8.** Labson D, et al. Presented at: ASH 67th Annual Meeting; December 6–9, 2025; Orlando, FL, USA. **9.** Challa DK, et al. *Curr Top Microbiol Immunol.* 2014;382:249–272. **10.** Ling LE, et al. *Clin Pharmacol Ther.* 2019;105(4):1031–1039. **11.** Seth NP, et al. *Mabs.* 2025;17(1):2461191. **12.** Yu F, et al. *Hum Vaccin Immunother.* 2026;22(1):2664331. **13.** Cossu M, et al. *Hum Vaccin Immunother.* 2025;21(1):2491269. **14.** Fattizzo B, et al. *HemaSphere* 2026;10(S1). **ACKNOWLEDGEMENTS:** We thank patients who participated in the trial and their families and caregivers, as well as investigators and staff who cared for patients and supported this trial and **ENERGY Investigators**: (Brazil) Jacques Kaufman, Andressa Lima Melo, Marcos Laercio Pontes Reis, Renato Tavares; (China) Rong Fu, Guangsheng He, Pengcheng He, Mei Hong, Zenghua Li, Miao Miao, Hongyan Tong, Shunqing Wang, Xiaoqin Wang, Chenlu Yang; (Czechia) Jiri Mayer; (Egypt) Raafat Abdel Fattah, Ashraf El Gandour, Mervat Mattar, Mohamed Moussa, Mohamed Samra; (France) Bernard Bonnotte, Clément Gourguechon; (Germany) Alexander Roth, Ingo Schwaner; (Greece) Argiris Symeonidis; (Hungary) Arpad Illes, Zsolt Lázár, Gabor Mikala, Zsolt Nagy; (Israel) Yariv Carasso, Chezi Ganzel, Ilya Kirgner, Itai Levi, Galia Spectre, Inna Tsoran-Rosenthal; (Italy) Bruno Fattizzo, Luana Fianchi, Andrea Patriarca; (Japan) Shunsuke Hatta, Masaki Iino, Koji Kawamura, Yoshitaka Miyakawa, Takashi Sonoki, Yasutaka Ueda; (Malaysia) Cheng Lay Teh, Sen Mui Tan; (Netherlands) Josephine Vos; (Poland) Jadwiga Dwilewicz-Trojaczek, Sebastian Grosicki, Tadeusz Robak; (South Korea) Jun Ho Jang, Hyunkyung Park; (Spain) Maria Aranzazu Alonso, Maria del Mar Urbano Ramos, Jose Angel Hernandez Rivas, Maria Eva Mingot Castellano, Gemma Moreno Jimenez, Irene Zamanillo; (United Kingdom) Quentin Hill, Sue Pavord, Syed Rashid Saeed Kazmi, David Simcox, Thomas Derrick Wayne; (United States) Gowthami Arepally, Troy Guthrie, Sung Ho Brian Lee, Darla Liles, Allyson Pishko. Medical writing support was provided by Chan Yoon, PharmD, of Lumanity Communications Inc., under the direction of the authors in accordance with Good Publication Practice guidelines (*Ann Intern Med.* 2022;175:1298-1304). Layout design for this poster was provided by Mugdha Rokade (SIRO Medical Writing Pvt. Ltd., India). This study and presentation were sponsored by Johnson & Johnson. **DISCLOSURES: BF** received honoraria for service on a consultancy/advisory board from Alexion, Johnson & Johnson, Novartis, Recordati, Roche, and Sobi. **IM** received research funding from Alexion, Bioverativ, Sanofi, Annexon, Incyte, Kezar, Principia, Rigel, and Novartis; and served in a consulting role for Alexion, Alpine/Vertex, Apellis, Johnson & Johnson, Novartis, Sanofi, and Recordati. **YU** received honoraria from Alexion, Chugai, Incyte, Kaken, Nippon Shinyaku, Novartis, Recordati, Sanofi, and Sobi; received consultancy fees from Alexion, Asahi Kasei, Chugai, Johnson & Johnson, Novartis, Ono, Recordati, Sanofi, and Sobi; received research funding from Chugai; and serves as an editor for the *International Journal of Hematology*. **DS, KS, MZ, TN, JC, JL, LL, SP,** and **CS** employees of Johnson & Johnson, may own stock/stock options in Johnson & Johnson. **PREVIOUSLY PRESENTED AT:** The European Hematology Association (EHA) Congress; June 11–14, 2026; Stockholm, Sweden.