Healthcare Resource Utilization Among Male Patients with Sjögren's Disease

Nora Sandorfi[1], Alexander Keenan[2], Elizabeth Adamson[2], Federico Zazzetti[2], Soumya D Chakravarty[2,3], Debbie Goldschmidt[4], Sakshi Sethi[4], Jessica Smith[4], Esen K Akpek[5]

[1]University of Pennsylvania, Philadelphia, PA, USA; [2]Johnson & Johnson, Horsham, PA, USA; [3]Drexel University School of Medicine, Philadelphia, PA, USA; [4]Analysis Group, New York, NY, USA; [5]Foster Center for Ocular Immunology and Inflammation, Duke University School of Medicine, Durham, NC, USA

Presenter: Archana Jeyakumar

Scan the QR code.

The QR code is intended to provide scientific information for individual reference, and the information should not be altered or reproduced in any way.

Key Takeaways

- **In this analysis, men with SjD demonstrated substantial clinical burden and HRU prior to diagnosis including inpatient admissions and outpatient and specialist visits, with further increases after diagnosis**
- **Men with SjD had more ophthalmology/optometry, vision care, ENT, or dental visits, whereas men with RA had more rheumatology or other joint or pain care visits**
- **The overall level of the burden for men with SjD was comparable to that for men with RA**
- **These findings are novel and highlight that there is a need for recognition of the burden and complex management of SjD in men**

Background

- Sjögren's disease (SjD) is a chronic systemic autoimmune disorder characterized by glandular dysfunction with manifestations ranging from oral/ocular dryness to life-threatening organ involvement[1]
- SjD predominantly affects women, but men may experience substantial clinical burden including higher rates of extraglandular manifestations that can lead to serious systemic complications[2,3]
- Evidence regarding healthcare resource utilization (HRU) in men with SjD is limited
- Comparing outcomes in men with SjD to those in men with rheumatoid arthritis (RA), a well-characterized systemic autoimmune disease, may provide context for understanding the burden associated with SjD

Objective

To compare clinical characteristics and HRU of male patients with SjD to a matched cohort of male patients with RA

Methods

Study design: retrospective observational matched cohort study

Study methods and analysis

- The index date was defined as the date of SjD or RA diagnosis
 - The baseline period was the 12 months before diagnosis
 - The follow-up period was the 12 months after diagnosis
- Patients were excluded from the RA cohort if they ever had an associated SjD diagnosis
- Patients with SjD were matched 1:1 to patients with RA by age
- Patient characteristics, diagnoses and symptoms, and HRU were assessed in the 12 months before and after diagnosis

US Veterans Health Administration data (January 1, 2015, to August 15, 2025)

- **Male adults (aged ≥18 years) with an initial clinical diagnosis of SjD on or after October 1, 2021**
- **Male adults (aged ≥18 years) with an initial clinical diagnosis of RA on or after October 1, 2021**

Additional eligibility criteria:

- A secondary (confirmatory) clinical diagnosis ≥30 days after the first clinical diagnosis
- ≥12 months' continuous enrollment before and after diagnosis
- Available dental data

HRU outcomes assessed:

- Inpatient admissions
- Emergency room admissions
- Outpatient visits
- Specialist visits and types
- Rheumatology or other joint or pain care visits
- Ophthalmology/optometry; other vision care; ear, nose, and throat (ENT); and dental visits

Results

- Overall, dry mouth, dry eyes, and ocular findings were more common among patients with SjD, whereas joint pain was more commonly observed for RA

Baseline characteristics of 617 male patients with SjD matched with 617 male patients with RA

Pre-Diagnosis Characteristics	Patients with SjD (n=617)	Patients with RA (n=617)
Demographics		
Age at diagnosis, years, mean (SD)	66.4 (13.4)	66.4 (13.4)
Race/ethnicity, n (%)		
White	339 (54.9)	389 (63.0)
Black or African American	135 (21.9)	117 (19.0)
Hispanic or Latino	76 (12.3)	44 (7.1)
Asian or Pacific Islander	21 (3.4)	16 (2.6)
Other or unknown	46 (7.5)	51 (8.3)
Disease Characteristics		
Charlson Comorbidity Index, mean (SD)	1.7 (1.9)	1.4 (1.9)
Number of relevant diagnoses/symptoms, mean (SD)	3.2 (2.0)	2.7 (1.8)
Most common diagnoses/symptoms before diagnosis,[a] n (%)		
Hypertension	416 (67.4)	399 (64.7)
Dry eyes	265 (42.9)	117 (19.0)
Joint pain	241 (39.1)	342 (55.4)
Depression	228 (37.0)	221 (35.8)
Chronic pain	120 (19.4)	123 (19.9)
Other autoimmune condition[b]	108 (17.5)	46 (7.5)
Dry mouth	106 (17.2)	18 (2.9)
Insomnia	100 (16.2)	87 (14.1)
Ocular findings[c]	86 (13.9)	27 (4.4)
Fatigue	80 (13.0)	84 (13.6)

[a]Most common diagnoses and symptoms defined as those occurring in >10% of patients within a cohort. [b]Other autoimmune conditions included RA, systemic lupus erythematosus, Hashimoto's thyroiditis, Grave's disease, scleroderma/systemic sclerosis, systemic vasculitis, psoriatic arthritis, inflammatory bowel syndrome, autoimmune hepatitis, myasthenia gravis, and cryoglobulinemia. [c]Ocular findings included sterile corneal ulcer/infiltration, corneal haze/scarring, corneal melt/perforation, uveitis, optic neuropathy/neuritis, episcleritis/scleritis, orbital inflammation, and papillary conjunctivitis. ***SD****=standard deviation.*

Patients with SjD and those with RA showed comparable HRU before diagnosis, and both groups experienced an increase in inpatient admissions after diagnosis

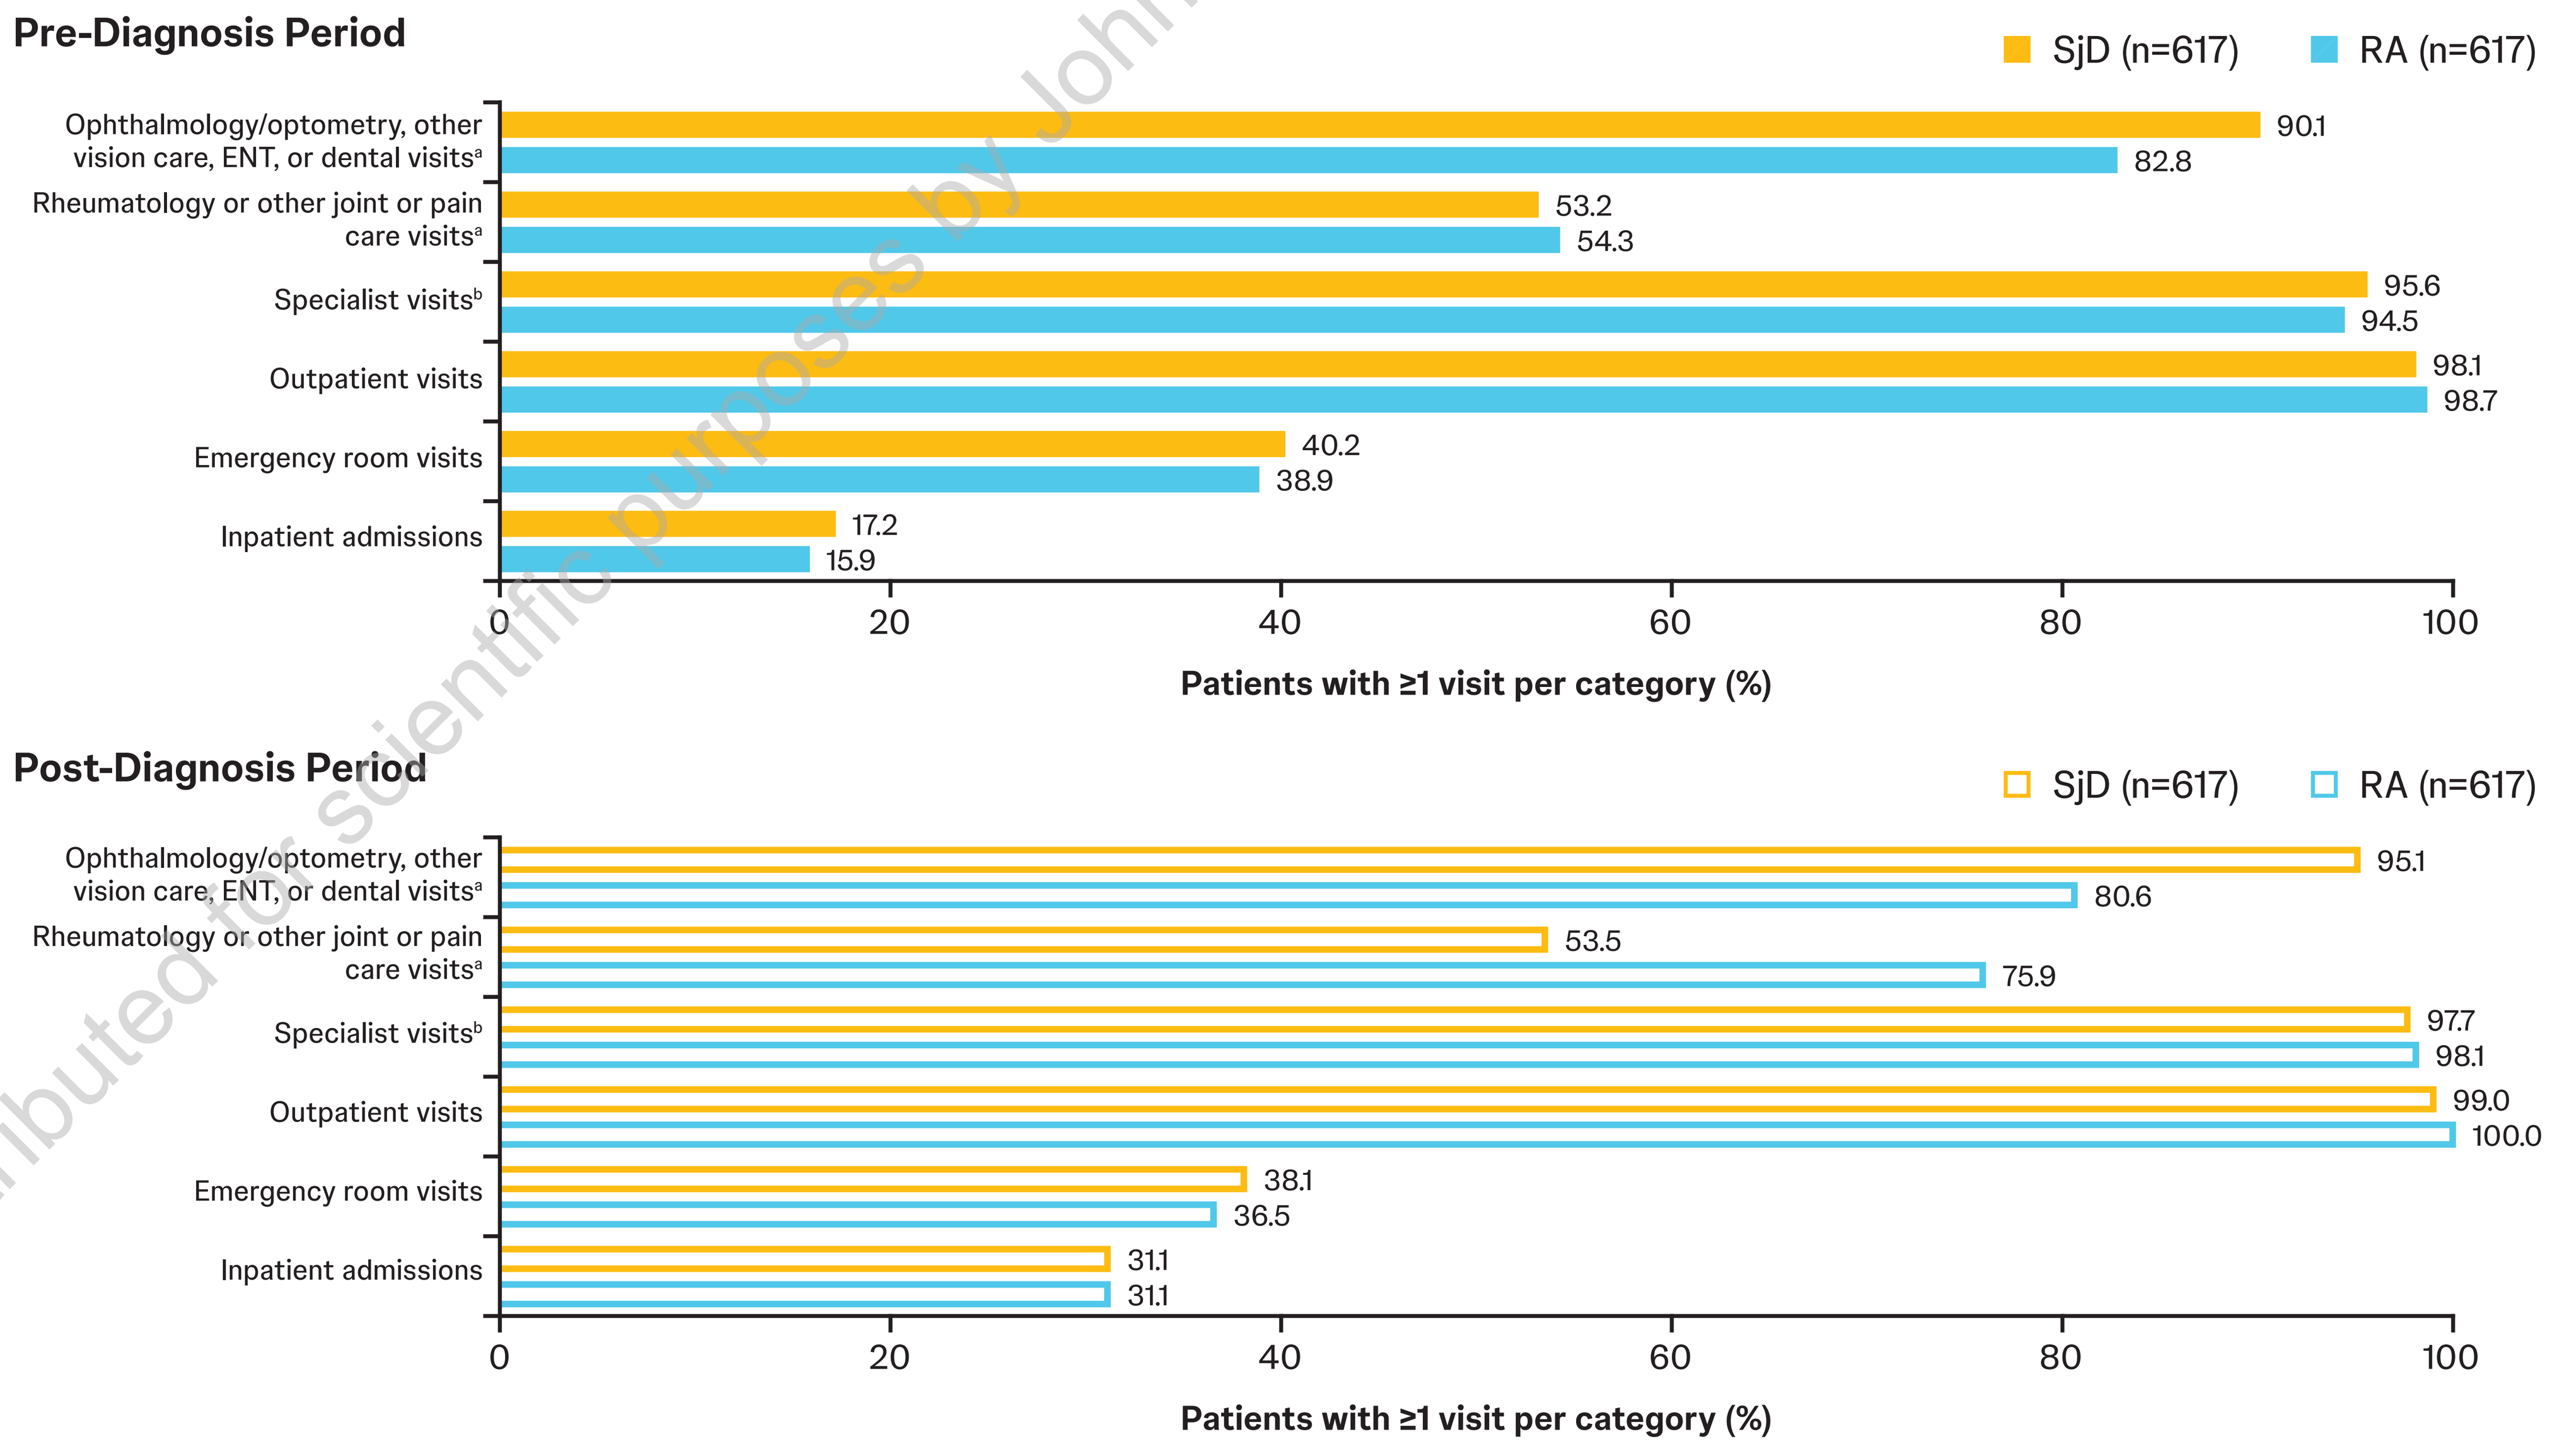
Pre-Diagnosis Period
SjD (n=617)
RA (n=617)
Ophthalmology/optometry, other vision care, ENT, or dental visits[a]
90.1
82.8
Rheumatology or other joint or pain care visits[a]
53.2
54.3
Specialist visits[b]
95.6
94.5
Outpatient visits
98.1
98.7
Emergency room visits
40.2
38.9
Inpatient admissions
17.2
15.9
0
20
40
60
80
100
Patients with ≥1 visit per category (%)
Post-Diagnosis Period
SjD (n=617)
RA (n=617)
Ophthalmology/optometry, other vision care, ENT, or dental visits[a]
95.1
80.6
Rheumatology or other joint or pain care visits[a]
53.5
75.9
Specialist visits[b]
97.7
98.1
Outpatient visits
99.0
100.0
Emergency room visits
38.1
36.5
Inpatient admissions
31.1
31.1
0
20
40
60
80
100
Patients with ≥1 visit per category (%)

[a]Patients with visits to more than one provider type were counted once in the combined categories. [b]Specialist visits included all types of outpatient care, excluding primary care and emergency room visits.

After diagnosis, patients with SjD and those with RA visited a similar number of specialist types per patient per year

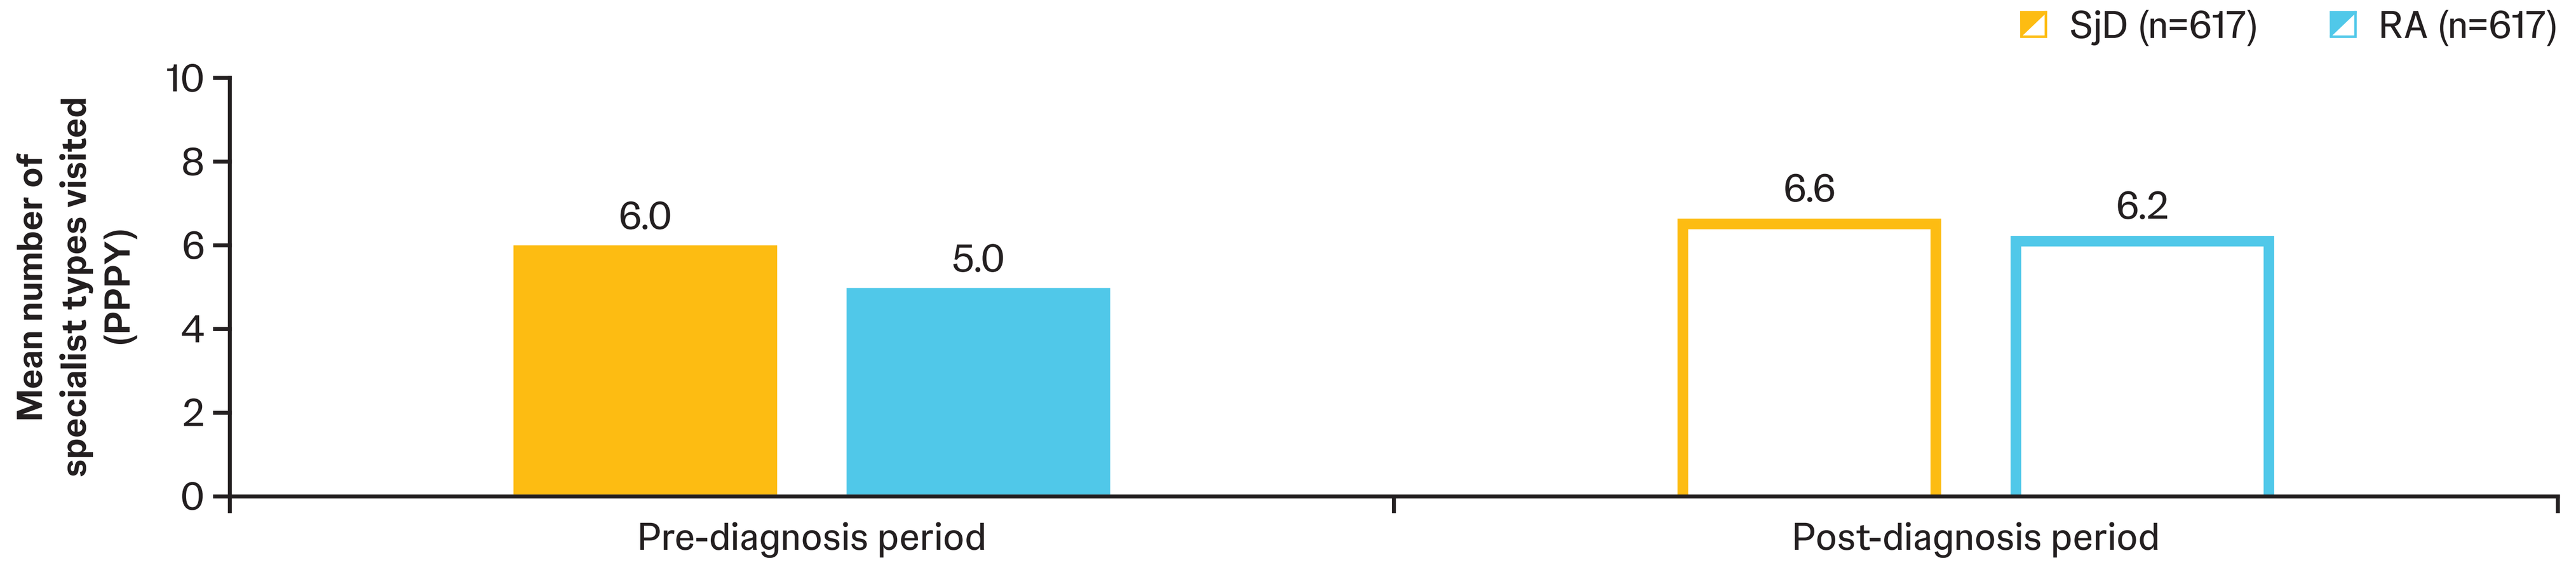
SjD (n=617)
RA (n=617)
Mean number of specialist types visited (PPPY)
10
8
6
4
2
0
6.0
5.0
6.6
6.2
Pre-diagnosis period
Post-diagnosis period

PPPY*=per patient per year.*

Patients with RA had a higher mean number of rheumatology or other joint or pain care visits after diagnosis, while patients with SjD had a higher mean number of vision, ENT, or dental visits

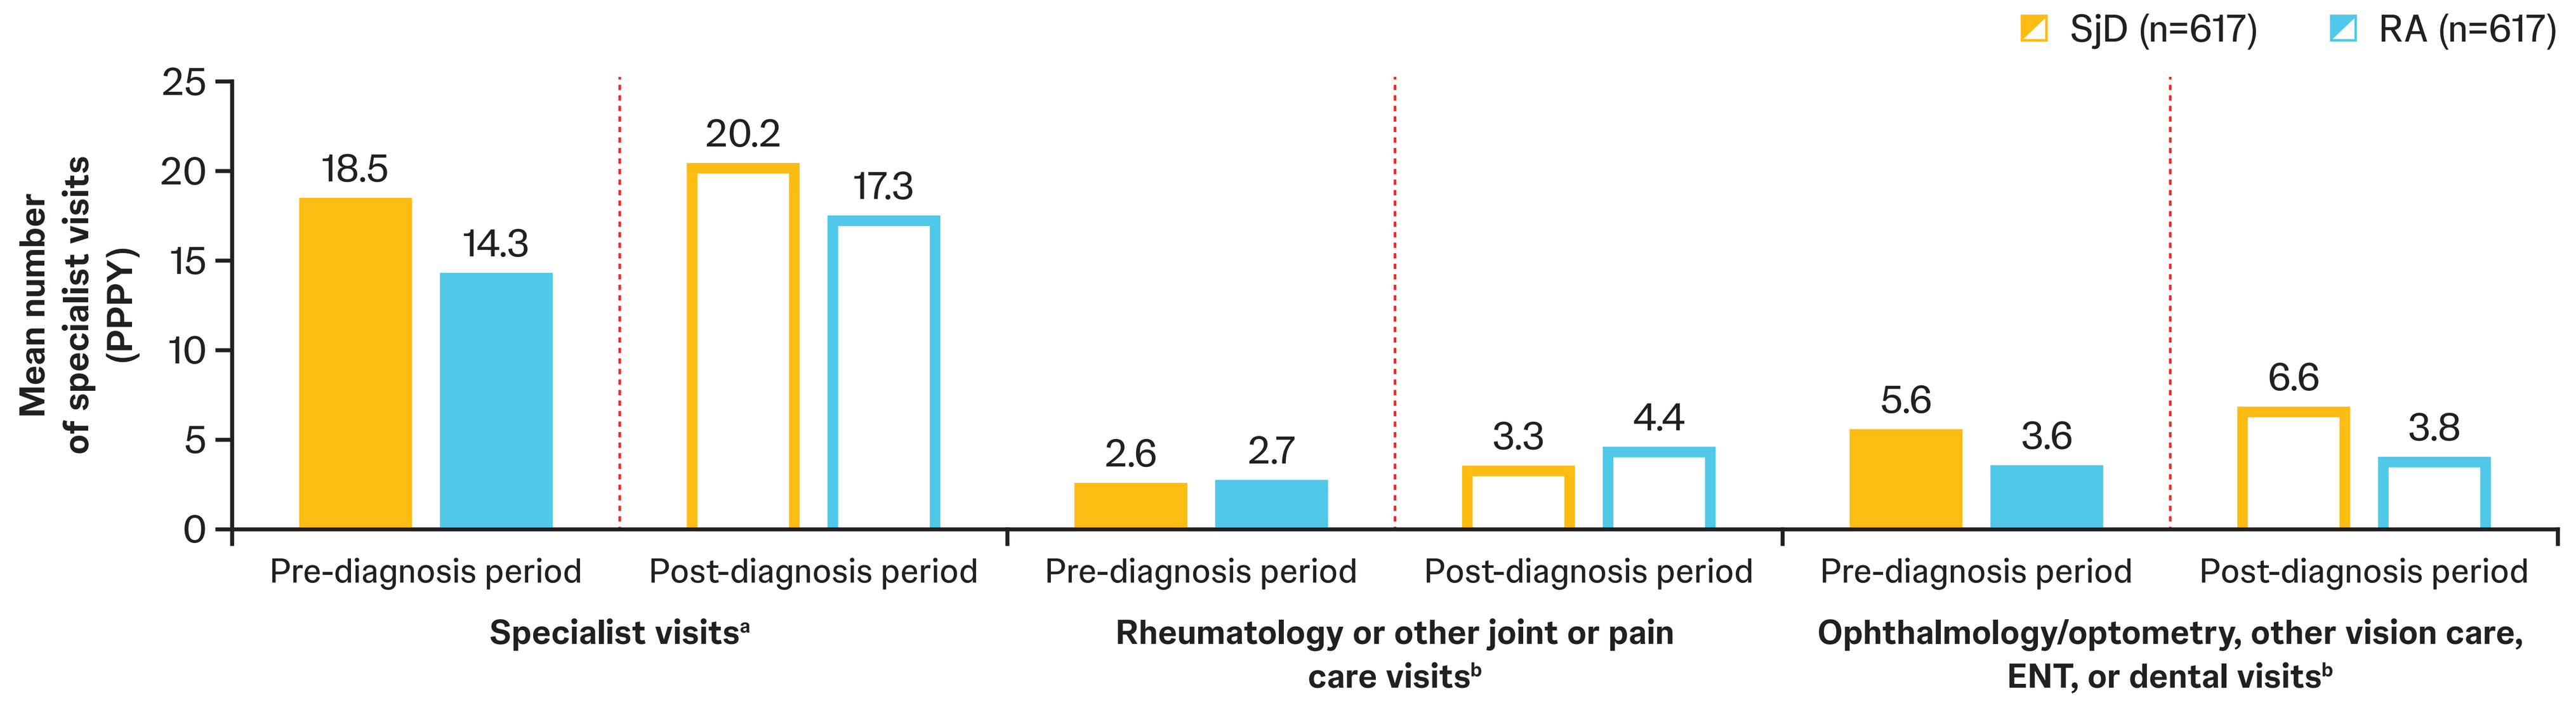
SjD (n=617)
RA (n=617)
Mean number of specialist visits (PPPY)
25
20
15
10
5
0
18.5
14.3
20.2
17.3
2.6
2.7
3.3
4.4
5.6
3.6
6.6
3.8
Pre-diagnosis period
Post-diagnosis period
Specialist visits[a]
Rheumatology or other joint or pain care visits[b]
Ophthalmology/optometry, other vision care, ENT, or dental visits[b]

[a]Specialist visits included all types of outpatient care, excluding primary care and emergency room visits. [b]Patients with visits to more than one provider type were counted once in the combined categories.

PRESENTED AT: The Congress of Clinical Rheumatology (CCR) – West 2026; Huntington Beach, CA, USA; September 17–20, 2026.
REFERENCES: 1. Beydon M, et al. *Nat Rev Rheumatol.* 2024;20:158–69. **2.** Virdee S, et al. *Clin Rheumatol.* 2017;36:2225–36. **3.** Mathews PM, et al. *Am J Ophthalmol.* 2015;160:447–52.
ACKNOWLEDGMENTS: This study was sponsored by Johnson & Johnson. Medical writing support was provided by Allison Michaelis on behalf of Twist Medical under the direction of the authors in accordance with Good Publication Practice guidelines (*Ann Intern Med.* 2022;175:1298–304). **DISCLOSURES: NS** has received consulting/advisory board fees from Bristol Myers Squibb, Johnson & Johnson, and Novartis. **AK**, **EA**, **FZ**, **SDC**, and **AJ** (presenter) are employees of and may own stock or stock options in Johnson & Johnson. **DG**, **SS**, and **JS** are employees of Analysis Group, Inc., a consulting company that has received research funding from Johnson & Johnson. **EKA** has received grant/research support from Dompé, the National Institutes of Health, Novartis, Sjögren's Foundation, and the US Department of Defense; and received consulting fees/honoraria from Alcon, Biotissue, Iolyx, Johnson & Johnson, Thea, and W.L. Gore & Associates.

This material is distributed for scientific purposes by Johnson & Johnson Innovative Medicine and is not for promotional use.