Guselkumab Efficacy on MDA Components and Structural Damage Progression in Participants With Active and Erosive Psoriatic Arthritis

Laura C. Coates,[1] Proton Rahman,[2] Philip J. Mease,[3,4] Oyediran Adelakun,[5] Soumya D. Chakravarty,[5,6] Alexa P. Kollmeier,[7] Weihong Hu,[8] Karissa Lozenski,[5] Frank R. Wellborne,[9] Arthur Kavanaugh,[10] Christopher T. Ritchlin[11]

[1]Nuffield Department of Orthopaedics, Rheumatology and Musculoskeletal Sciences, University of Oxford, Botnar Research Centre Oxford, UK; [2]Faculty of Medicine, Division of Rheumatology, Memorial University of Newfoundland, St. Johns, NL, Canada; [3]Rheumatology Research, Providence Swedish Medical Center, Seattle, WA, USA; [4]University of Washington School of Medicine, Seattle, WA, USA; [5]Johnson & Johnson, Horsham, PA, USA; [6]Drexel University College of Medicine, Philadelphia, PA, USA; [7]Johnson & Johnson, San Diego, CA, USA; [8]Johnson & Johnson, Spring House, PA, USA; [9]Director, Rheumatic Innovative Therapies, Houston Institute of Clinical Research, Houston, TX, USA; [10]Center for Innovative Therapy, Division of Rheumatology, Allergy, Immunology, University of California San Diego, La Jolla, CA, USA; [11]Department of Medicine, Allergy/Immunology and Rheumatology, University of Rochester Medical Center, Rochester, NY, USA

Scan the QR code.
The QR code is intended to provide scientific information for individual reference, and the information should not be altered or reproduced in any way.

Key Takeaways

- **In APEX, GUS-treated pts had numerically greater response rates for achieving individual MDA components and resolution of SJC/TJC vs PBO pts at W24 with continued improvement through W48**
- **Achievement of MDA criteria, as early as W12, was associated with reduced structural damage progression at W48 for both GUS dosing regimens**

Background

- Guselkumab (GUS) is a fully human, dual-acting[1] monoclonal antibody that selectively inhibits the interleukin-23p19 subunit and is approved for patients with active psoriatic arthritis (PsA)
- In APEX, GUS demonstrated multidomain efficacy and significantly inhibited structural damage progression vs placebo (PBO) at Week (W) 24 in participants (pts) with active & erosive PsA[2]
- Minimal disease activity (MDA) is a composite measure that evaluates low levels of disease activity across multiple PsA domains: peripheral joint activity, psoriatic skin disease, pain, patient global assessment, physical function, and health-related quality of life[3]

Objective

Evaluate the efficacy of GUS in achieving individual components of MDA and their association with structural damage progression through W48

Methods

Inclusion Criteria

- ✓ Biologic-naive
- ✓ Age ≥18 years
- ✓ Active PsA ≥6 months (despite prior csDMARD, apremilast, NSAID); CASPAR criteria met
- ✓ ≥3 SJC; ≥3 TJC; CRP ≥0.3 mg/dL
- ✓ **≥2 erosive joints on hand/foot radiographs**

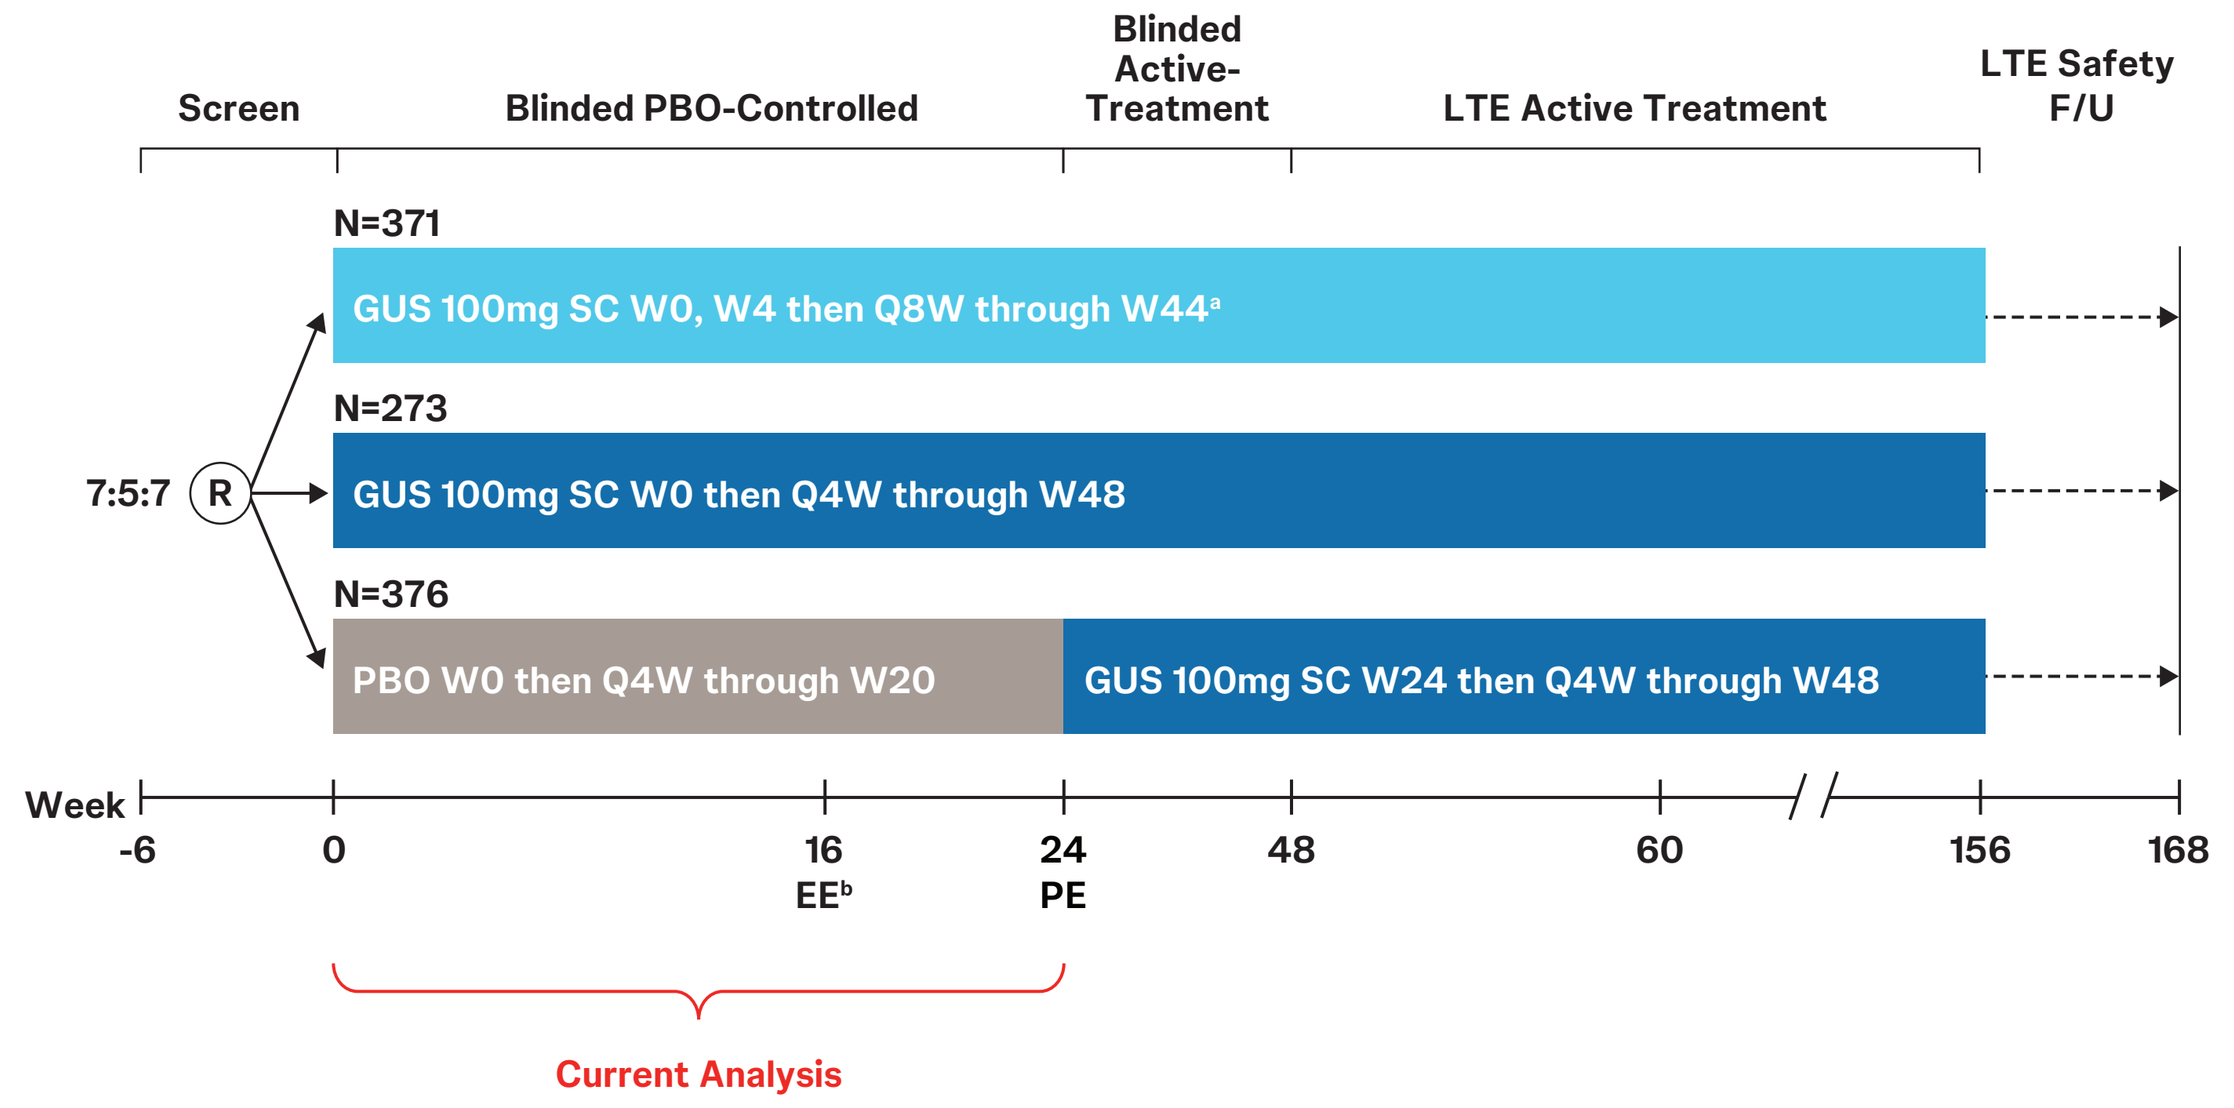

Screen
Blinded PBO-Controlled
Blinded Active-Treatment
LTE Active Treatment
LTE Safety F/U
N=371
GUS 100mg SC W0, W4 then Q8W through W44ᵃ
7:5:7 R
N=273
GUS 100mg SC W0 then Q4W through W48
N=376
PBO W0 then Q4W through W20
GUS 100mg SC W24 then Q4W through W48
Week -6 0 16 24 48 60 156 168
EEᵇ
PE
Current Analysis

Endpoints

The proportion of pts achieving the following outcomes through W48:

- Individual MDA components: SJC ≤1, TJC ≤1, Pt Pain ≤15, PASI ≤1, PtGA ≤20, HAQ-DI ≤0.5, LEI ≤1
- SJC=0
- TJC=0

LSM changes in PsA modified vdH-S score at W24 and W48 among GUS-randomized pts were analyzed by W12, W24, and W48 response status (achievement of individual MDA components and SJC=0/TJC=0)

Statistical Analyses

- NRI was used for:
 - Pts who initiated/increased csDMARDs or oral corticosteroids or initiated protocol-prohibited medication/therapies through W24
 - Pts who discontinued study intervention for any reason other than ND/MD through W48
- Data affect by ND/MD were not used
- Other missing data were accounted in the model (GLMM), followed by NRI
- Response rates for each outcome were assessed in pts with a baseline score greater than the relative response threshold
- For change in vdH-S scores, data affected by ND/MD were not used; missing data were imputed using MI

*ᵃPBO SC W8 then Q8W through W48 administered to maintain blinding. ᵇEE if <20% improvement from baseline in both TJC and SJC at W16. EE pts may initiate/increase dose of permitted medication up to the maximum dose, at the investigator's discretion. **CASPAR**=ClASsification criteria for Psoriatic ARthritis, **CRP**=C-reactive protein, **csDMARD**=conventional synthetic disease modifying antirheumatic drug, **EE**=early escape, **F/U**=follow-up, **GLMM**=Generalized Linear Mixed Model, **HAQ-DI**=Health Assessment Questionnaire-Disability Index, **LEI**=Leeds Enthesitis Index (tender entheseal points), **LSM**=least squares mean, **LTE**=long-term extension, **MD**=major disruption (Ukraine/Russia crisis), **MI**=multiple imputation, **ND**=natural disaster (COVID-19 site access restrictions), **NRI**=nonresponder imputation, **NSAID**=nonsteroidal anti-inflammatory drug, **PASI**=Psoriasis Area and Severity Index, **PE**=primary endpoint, **Pt Pain**=Patient-reported Pain, **PtGA**=Patient Global Assessment of disease activity (arthritis and psoriasis), **Q4W/Q8W**=every 4/8 weeks, **R**=randomization, **SC**=subcutaneous, **SJC**=swollen joint count, **TJC**=tender joint count, **vdH-S**=van der Heijde-Sharp.*

Results

Baseline demographics were well balanced among treatment groups

- Mean baseline disease characteristics were consistent across treatment groups

	GUS Q4W (N=273)	GUS Q8W (N=371)	PBO (N=376)	Total (N=1020)
Demographics				
Age, yrs	52.2 (13.2)	53.2 (12.9)	53.5 (13.0)	53.0 (13.0)
Male	55%	54%	57%	55%
White	85%	82%	83%	83%
BMI, kg/m²	29.4 (6.0)	29.0 (5.6)	28.9 (5.7)	29.1 (5.7)
PsA Characteristics				
PsA Disease Duration, yrs	7.5 (7.1)	7.2 (7.6)	7.2 (6.9)	7.3 (7.2)
SJC (0-66)	11.6 (9.4)	12.1 (8.5)	11.8 (8.9)	11.9 (8.9)
TJC (0-68)	21.2 (14.6)	20.6 (13.4)	20.5 (13.9)	20.7 (13.9)
PASI (0-72)	7.6 (8.3)[a]	8.3 (10.1)[b]	8.2 (9.5)[c]	8.1 (9.4)[d]
Pt Pain (VAS; 0-10 cm)	5.9 (2.2)	5.9 (2.1)	5.9 (2.1)	5.9 (2.1)
PtGA (VAS; 0-10 cm)	5.9 (2.2)	5.9 (2.1)	5.9 (2.0)	5.9 (2.1)
HAQ-DI (0-3)	1.2 (0.7)	1.2 (0.6)	1.2 (0.7)	1.2 (0.7)
LEI (1-6)	3.2 (1.8)[e]	3.0 (1.7)[f]	3.0 (1.6)[g]	3.1 (1.7)[h]
CRP, mg/dL	1.7 (2.9)	1.5 (2.0)	1.7 (2.5)	1.6 (2.5)

*[a]N=269, [b]N=364, [c]N=369, [d]N=1002, [e]N=271, [f]N=365, [g]N=372, [h]N=1008. Data shown are mean (SD) unless otherwise noted. **BMI**=body mass index, **VAS**=visual analog scale.*

Response rates were greater for GUS-randomized pts compared to PBO-randomized pts at W24

- The separation of response rates for GUS-randomized vs PBO-randomized pts occurred as early as W8 (nominal p<0.05)
- Response rates continued to increase through W48

Proportions of Pts Achieving Individual MDA Components and SJC=0/TJC=0 Through W48

GUS Q4W

Week	4	8	12	16	20	24	28	36	48
SJC ≤1	7.5	19.0	26.9‡	35.4†	41.7*	47.6*	51.5	54.1	59.0
TJC ≤1	1.7	5.1	9.3‡	12.1	15.9‡	19.1†	22.7	24.1	28.0
PASI ≤1	13.4	N/A	46.4*	N/A	N/A	71.2*	81.5	N/A	83.4
Pt Pain ≤15	4.6	11.1	18.1	23.9	28.8†	29.5†	29.9	34.9	35.3
PtGA ≤20	8.5	17.0‡	25.5†	30.5†	33.8*	40.5*	42.1	44.6	46.8
HAQ-DI ≤0.5	12.7‡	15.9	16.3	21.0	23.3	26.5‡	28.2	30.1	32.8
LEI ≤1	39.9‡	N/A	54.0	N/A	N/A	58.4	66.6	66.6	65.0
SJC=0	3.8	13.6‡	18.5‡	24.0‡	28.7†	36.7*	39.8	44.2	48.3
TJC=0	1.3	2.2	4.8	6.5	9.8‡	12.1†	15.8	15.1	21.4

GUS Q8W

Week	4	8	12	16	20	24	28	36	48
SJC ≤1	7.9	19.0‡	25.8‡	32.5‡	38.7†	43.5*	46.4	51.1	53.0
TJC ≤1	1.5	5.9	8.5	11.8	13.7	17.4†	21.2	22.2	29.2
PASI ≤1	13.0	N/A	47.4*	N/A	N/A	63.5*	70.6	N/A	76.3
Pt Pain ≤15	6.7	15.1‡	19.7‡	25.1‡	29.7*	30.0†	34.0	38.8	42.1
PtGA ≤20	7.9	18.7†	22.7†	33.0*	36.2*	37.1*	43.9	45.9	48.9
HAQ-DI ≤0.5	12.5†	11.5	21.2	26.6†	29.0*	28.0†	32.6	34.6	35.9
LEI ≤1	37.8	N/A	52.8	N/A	N/A	60.1	56.4	61.4	68.2
SJC=0	4.4	12.2‡	18.4‡	24.5†	30.5*	32.2*	36.2	42.4	44.0
TJC=0	0.7	1.9	3.4	6.8	7.5	12.0†	14.5	14.8	18.6

PBO (W4–W24); PBO→GUS Q4W (W28–W48)

Week	4	8	12	16	20	24	28	36	48
SJC ≤1	5.5	13.8	19.0	24.7	28.1	28.1	34.2	43.0	50.0
TJC ≤1	2.0	3.5	5.0	8.3	9.3	10.1	11.6	17.4	22.4
PASI ≤1	9.1	N/A	19.6	N/A	N/A	27.5	36.8	N/A	74.2
Pt Pain ≤15	3.8	9.7	13.5	17.8	17.5	19.5	24.1	33.5	40.4
PtGA ≤20	6.8	10.5	14.8	19.8	20.9	21.2	29.1	42.7	47.0
HAQ-DI ≤0.5	6.2	10.1	17.3	17.8	17.1	18.7	22.0	28.0	31.4
LEI ≤1	28.6	N/A	46.8	N/A	N/A	53.3	63.1	66.2	70.2
SJC=0	3.3	7.4	11.7	15.9	18.8	19.3	22.8	35.1	38.5
TJC=0	0.5	1.4	2.6	5.5	5.5	5.5	6.2	11.0	13.2

0% 20% 40% 60% 80% 100%

*Nominal *p<0.001, †p<0.01, ‡p<0.05 vs PBO. P-values are based on GLMM, are considered nominal, and may not be used to claim statistical significance. **N/A**=not available; the assessment was not collected at this time point.*

Among GUS-randomized pts, pts achieving response at W24 or W48 had less structural damage progression compared with nonresponders across endpoints

LSM Change in vdH-S at W24 by W24 Responders/Nonresponders

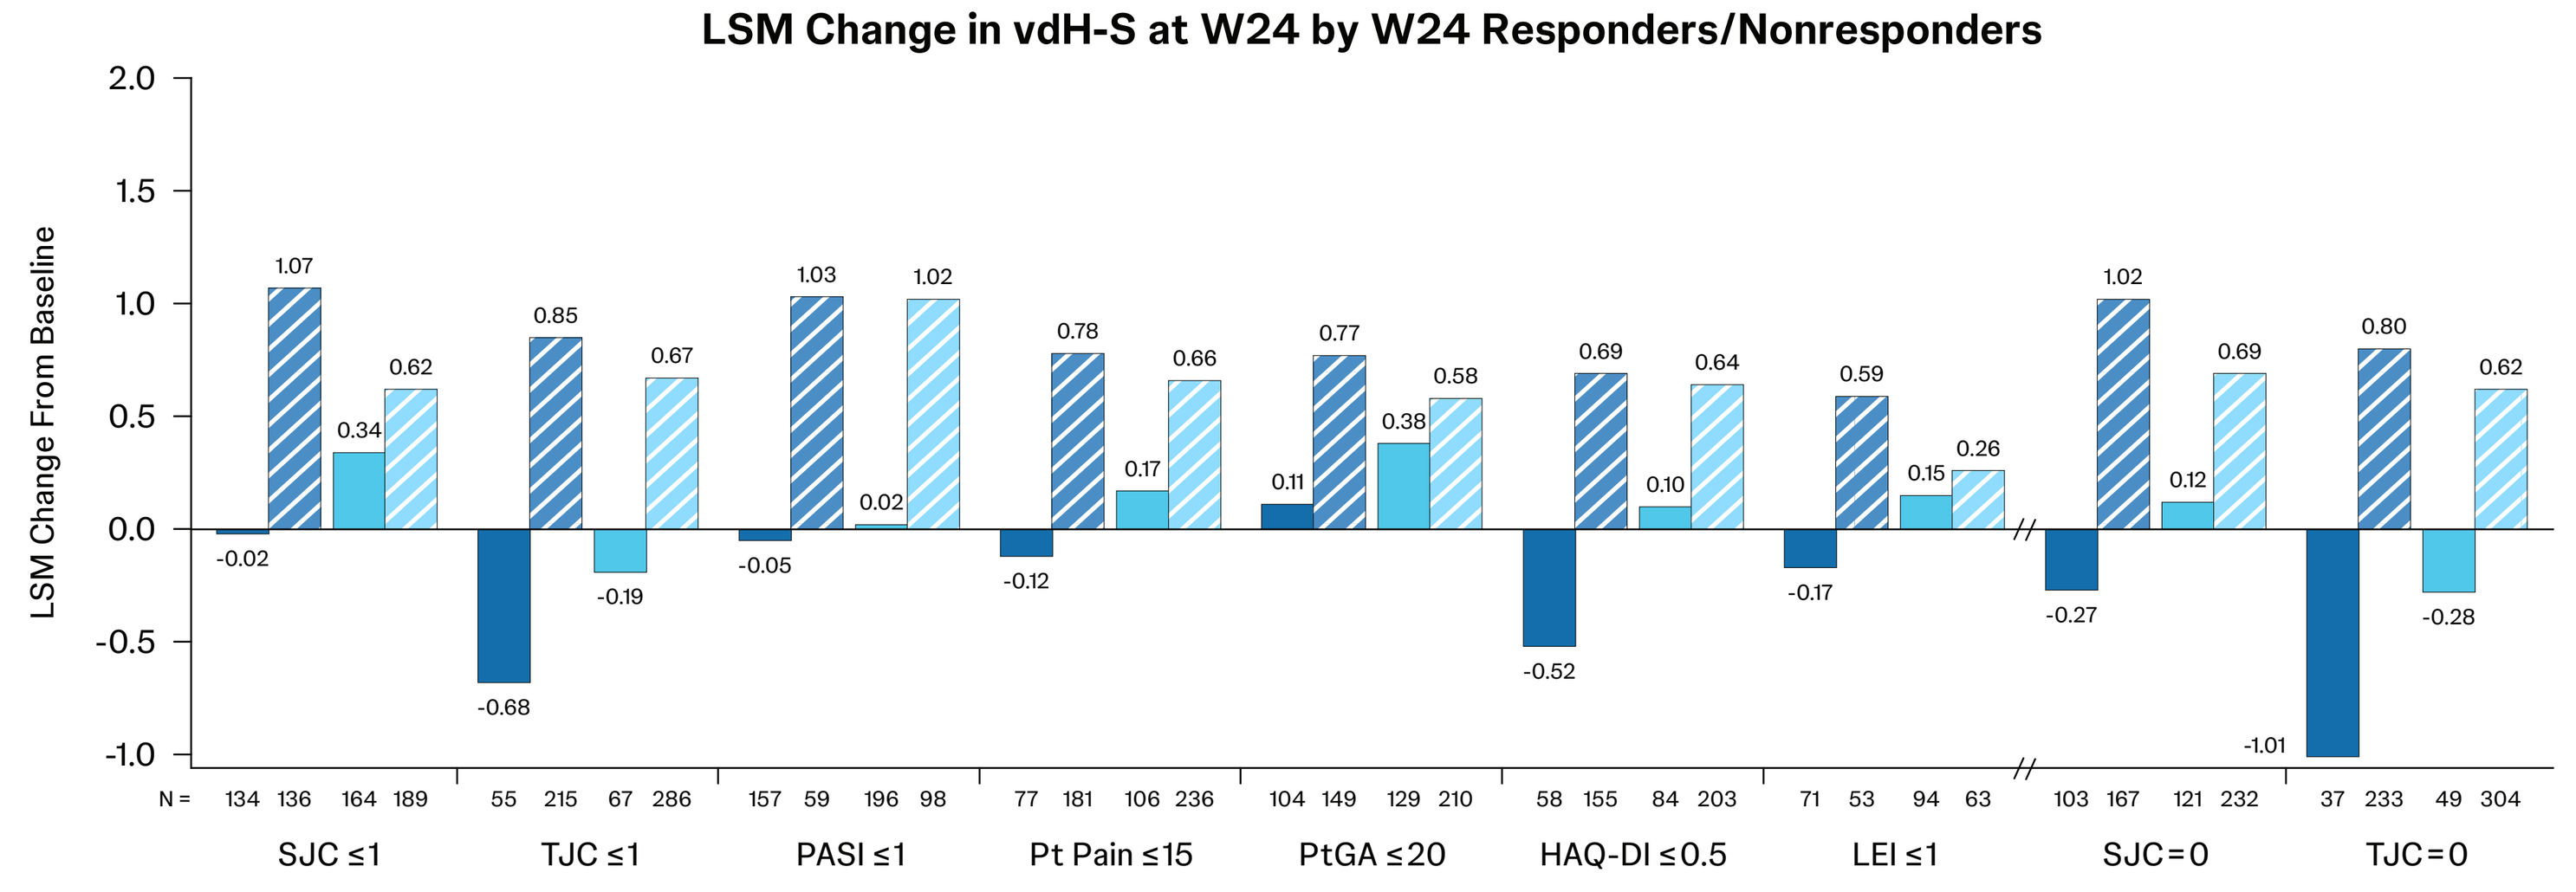

LSM Change From Baseline
SJC ≤1
TJC ≤1
PASI ≤1
Pt Pain ≤15
PtGA ≤20
HAQ-DI ≤0.5
LEI ≤1
SJC=0
TJC=0

LSM Change in vdH-S at W48 by W48 Responders/Nonresponders

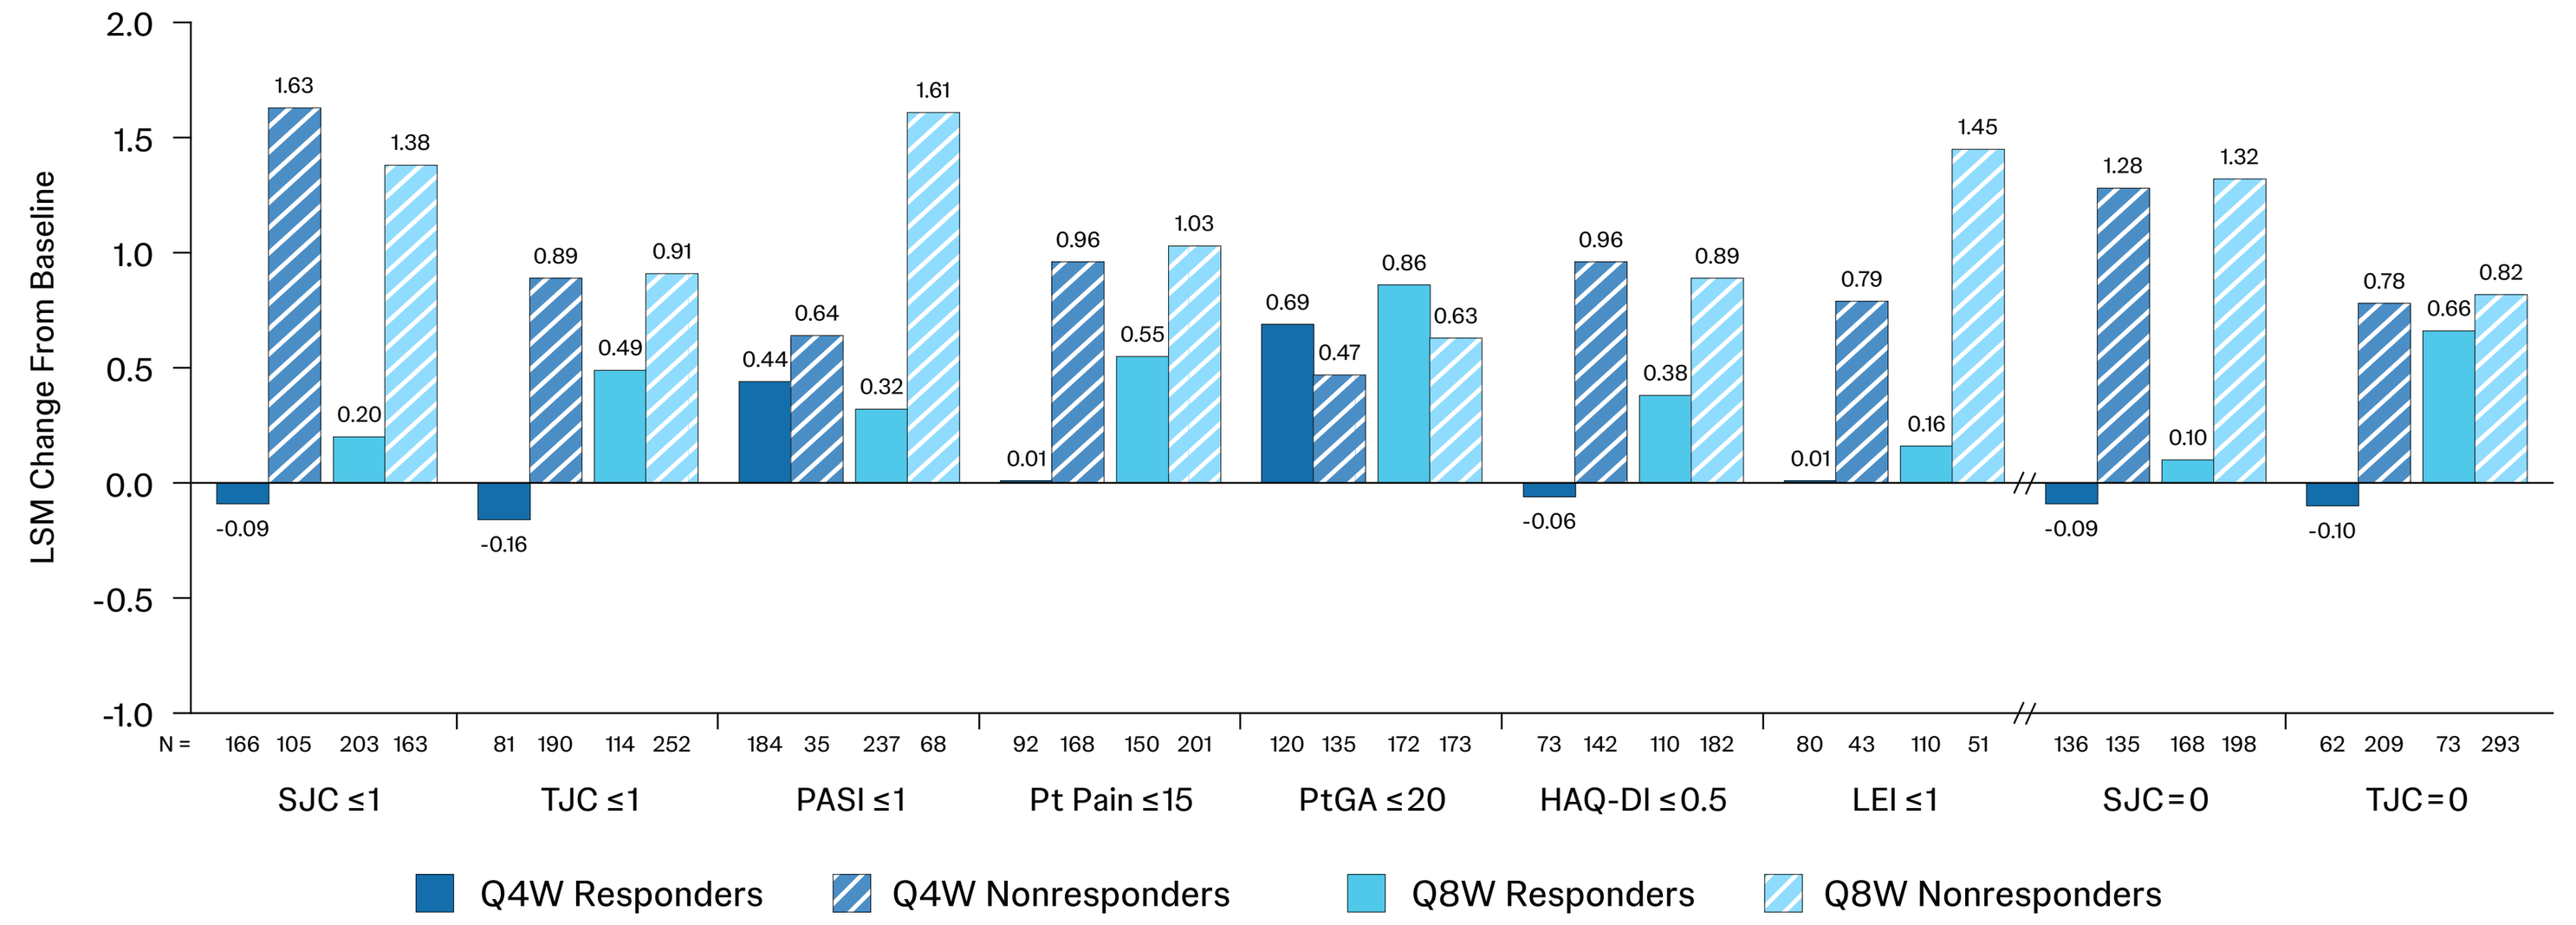

LSM Change From Baseline
SJC ≤1
TJC ≤1
PASI ≤1
Pt Pain ≤15
PtGA ≤20
HAQ-DI ≤0.5
LEI ≤1
SJC=0
TJC=0
Q4W Responders
Q4W Nonresponders
Q8W Responders
Q8W Nonresponders

Among GUS-randomized pts, clinical response at W12 or W24 was associated with less structural damage progression at W48 compared with nonresponders

LSM Change in vdH-S at W48 by W12 Responders/Nonresponders

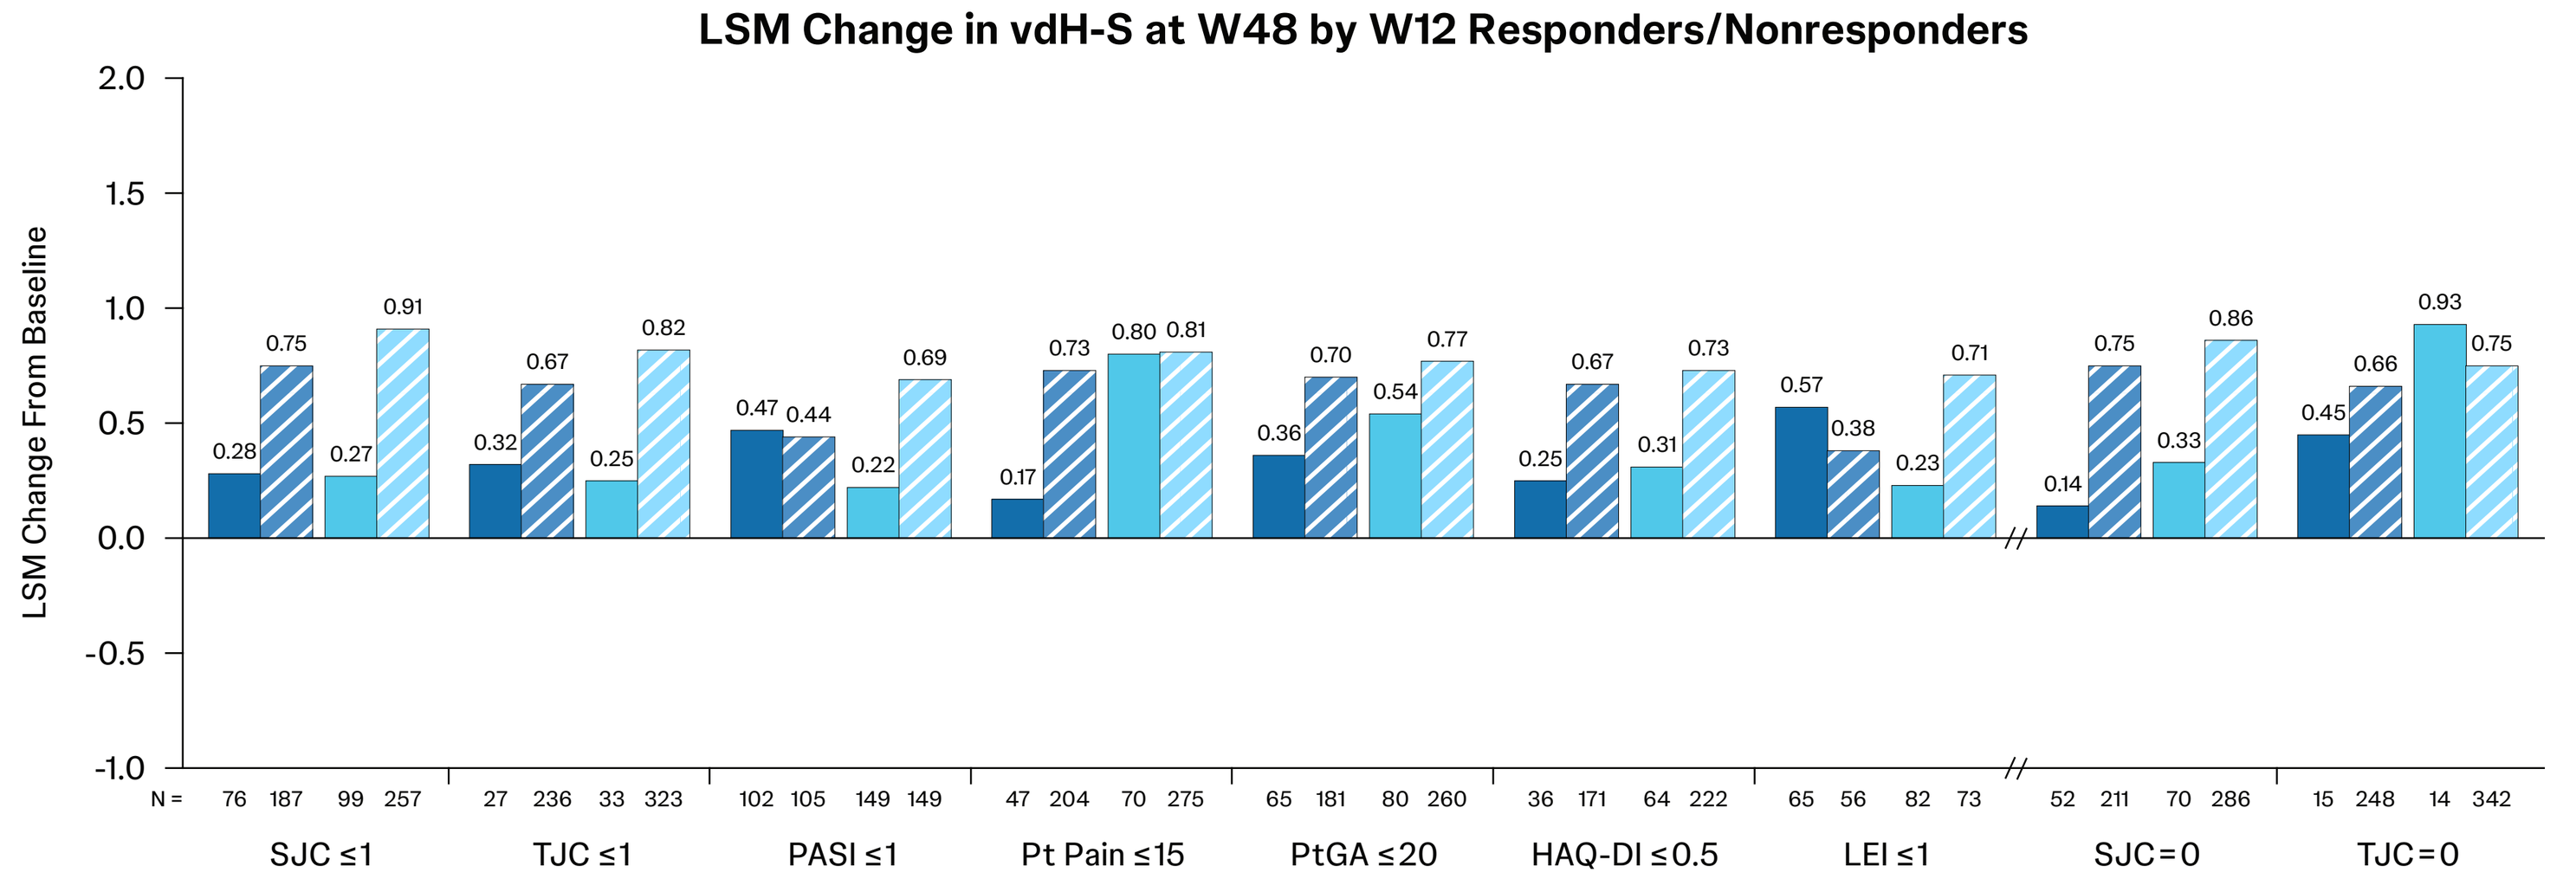

LSM Change From Baseline
SJC ≤1
TJC ≤1
PASI ≤1
Pt Pain ≤15
PtGA ≤20
HAQ-DI ≤0.5
LEI ≤1
SJC=0
TJC=0

LSM Change in vdH-S at W48 by W24 Responders/Nonresponders

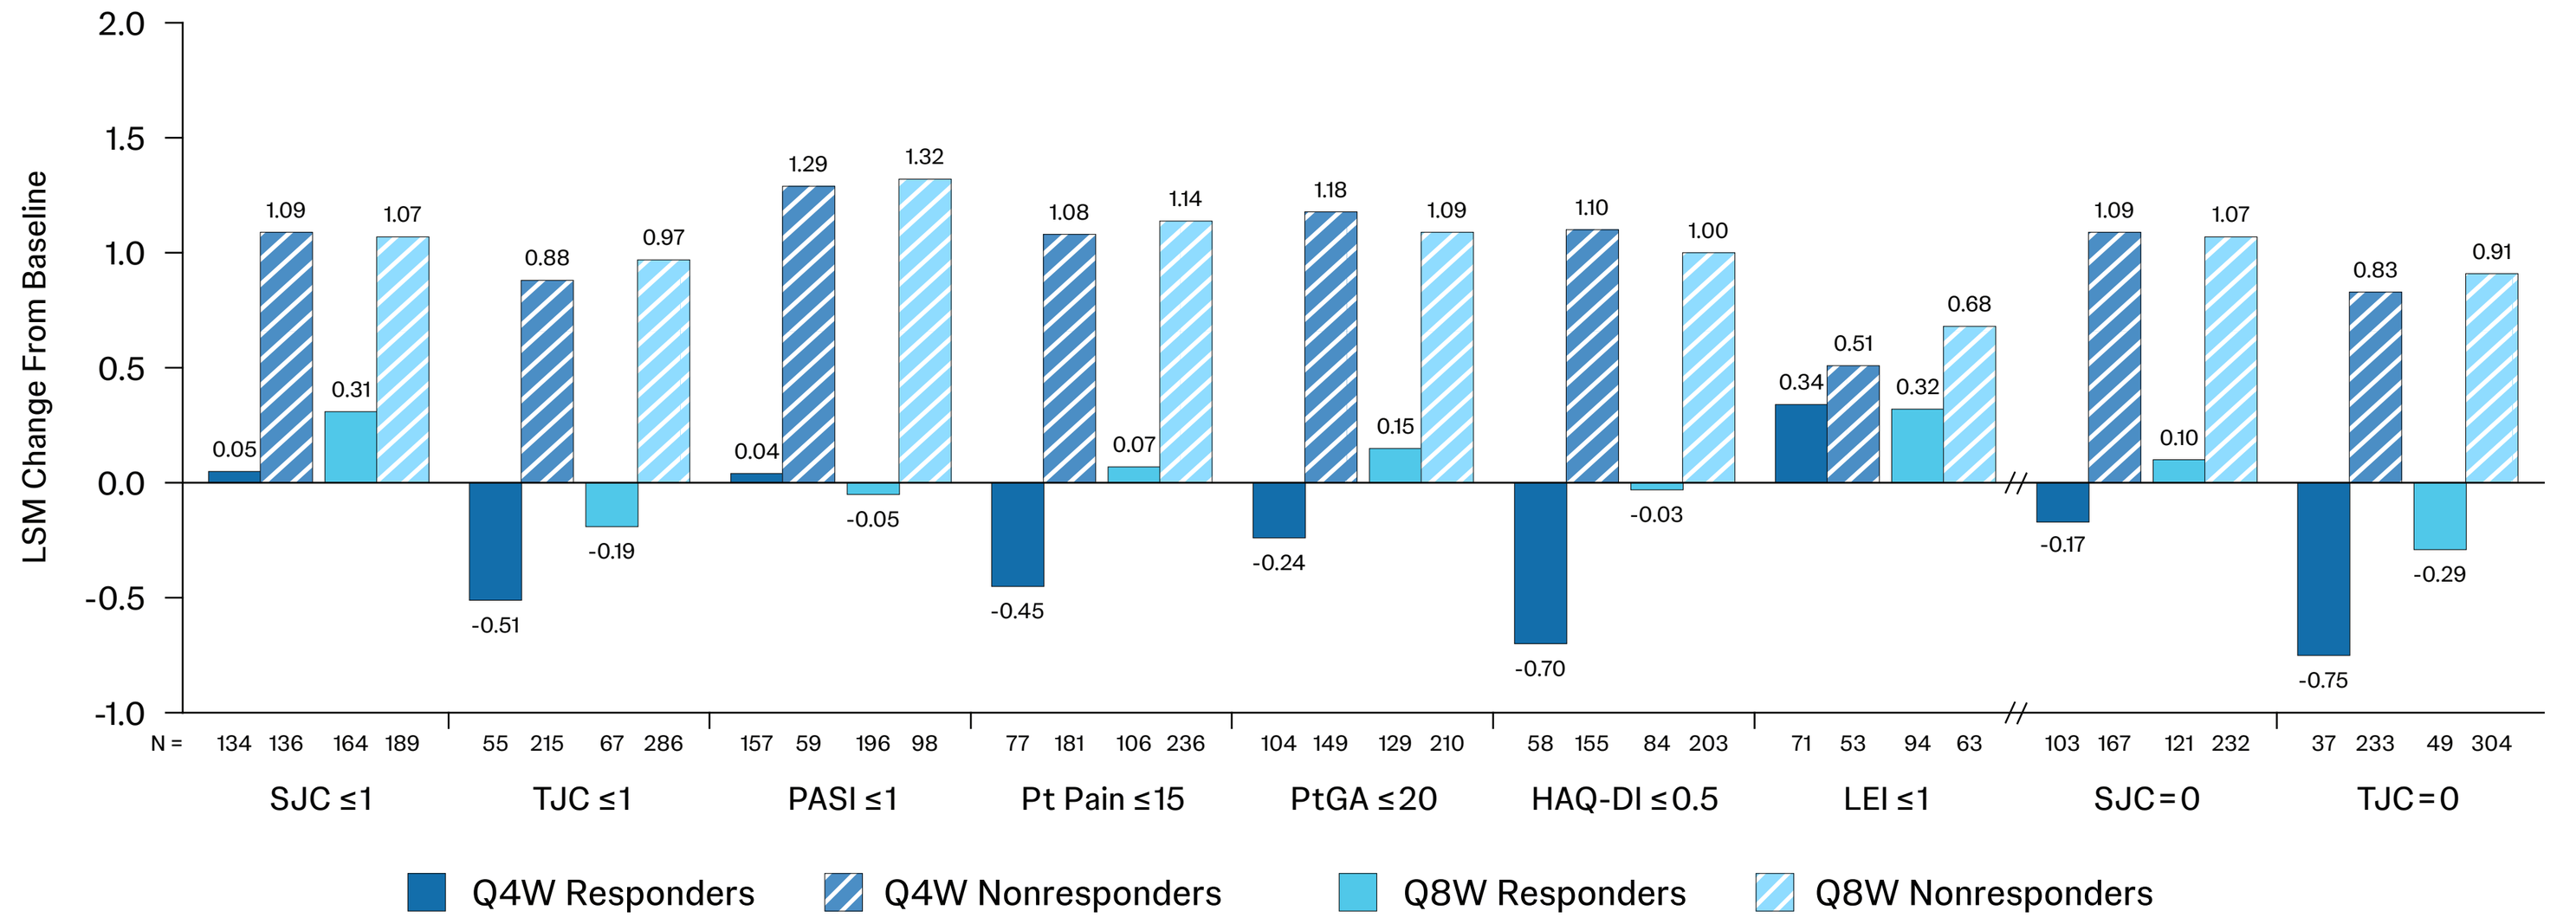

LSM Change From Baseline
SJC ≤1
TJC ≤1
PASI ≤1
Pt Pain ≤15
PtGA ≤20
HAQ-DI ≤0.5
LEI ≤1
SJC=0
TJC=0
Q4W Responders
Q4W Nonresponders
Q8W Responders
Q8W Nonresponders

PRESENTED AT: Congress of Clinical Rheumatology-West 2026; September 17-20, 2026; Huntington Beach, CA.

REFERENCES: 1. Sachen KL, et al. *Front Immunol.* 2025;16:1532852. **2.** Mease PJ, et al. *Ann Rheum Dis.* 2025;84:1983-94. **3.** Coates LC, et al. *Ann Rheum Dis.* 2010;69:48-53.

ACKNOWLEDGMENTS: Medical writing support was provided by Alyssa Cortina, Pharm D, under the direction of the authors in accordance with Good Publication Practice guidelines (*Ann Intern Med.* 2022;175:1298-1304). Sponsored by Johnson & Johnson.

DISCLOSURES: LCC has received grants/research support from AbbVie, Amgen, Celgene, Eli Lilly, Johnson & Johnson, Novartis, Pfizer, and UCB; has worked as a paid consultant for AbbVie, Amgen, Bristol Myers Squibb, Celgene, Eli Lilly, Galapagos, Gilead, Johnson & Johnson, Moonlake, Novartis, Pfizer, and UCB; and has been paid as a speaker for AbbVie, Amgen, Biogen, Celgene, Eli Lilly, Galapagos, Gilead, GlaxoSmithKline, Johnson & Johnson, Medac, Novartis, Pfizer, and UCB. **PR** has received consulting fees from AbbVie, Amgen, Bristol Myers Squibb, Celgene, Eli Lilly, Johnson & Johnson, Merck, Novartis, Pfizer, and UCB; has received meeting attendance/travel support from Johnson & Johnson; and has received research grants from Johnson & Johnson and Novartis. **PJM** has received consulting fees from AbbVie, Amgen, Bristol Myers Squibb, Century, Cullinan, Eli Lilly, Inmagene, Johnson & Johnson, Merck, Moonlake, Novartis, Pfizer, Spyre, Sun Pharma, Takeda, and UCB; has received grants from AbbVie, Amgen, Bristol Myers Squibb, Eli Lilly, Johnson & Johnson, Moonlake, Novartis, Sana, Takeda, and UCB; and has received speaker fees from AbbVie, Amgen, Eli Lilly, Johnson & Johnson, Novartis, and UCB. **OA, SDC, APK,** and **KL** are employees of Johnson & Johnson, and may own stock or stock options in Johnson & Johnson. **WH** is an employee of Cytel, Inc. providing statistical support (funded by Johnson & Johnson). **FRW** has received clinical research support from AbbVie, Bristol Myers Squibb, Johnson & Johnson, Novartis, and Pfizer; has received consulting fees from AbbVie, Bristol Myers Squibb, Eli Lilly, Johnson & Johnson, Novartis, and UCB; and has been a paid speaker for AbbVie, Bristol Myers Squibb, Johnson & Johnson, and UCB. **AK** has received consulting fees from AbbVie, Amgen, Bristol Myers Squibb, Eli Lilly, Johnson & Johnson, Moonlake, Novartis, Pfizer, and UCB. **CTR** has been a consultant for AbbVie, Eli Lilly, Johnson & Johnson, Novartis, and UCB; and has received grant/research support from Johnson & Johnson, Novartis, and UCB.

This material is distributed for scientific purposes by Johnson & Johnson Innovative Medicine and is not for promotional use.