Clinically Relevant Anti-Vaccine and Virus Antibodies in Patients With Sjögren's Disease Treated With Nipocalimab: Post-hoc Analysis of the DAHLIAS Study

Faye Yu,[1] Eugene Myshkin,[1] Jonathan J. Hubbard,[2] Kim Campbell,[2] Matthew J. Loza,[2] Kathleen A. Packard,[3] Dessislava Dimitrova,[2] Carolyn Cuff,[1] Sheng Gao[2]

[1]Johnson & Johnson, Cambridge, MA, USA; [2]Johnson & Johnson, Spring House, PA, USA; [3]Johnson & Johnson, Horsham, PA, USA

Scan the QR code.
The QR code is intended to provide scientific information for individual reference, and the information should not be altered or reproduced in any way.

This material is distributed for scientific purposes by Johnson & Johnson Innovative Medicine and is not for promotional use

Key Takeaways

- Nipocalimab-treated participants with Sjögren's Disease elicited IgG responses to SARS-CoV-2 vaccination and/or infection
- Nipocalimab reduced pre-existing anti-vaccine antibodies to a similar extent as total IgG, consistent with the mechanism of action of nipocalimab; most nipocalimab-treated participants who were immune to TT and VZV at baseline maintained protective IgG levels during and after treatment
- Total and vaccine-specific IgG returned to baseline levels after treatment cessation
- Results suggest that nipocalimab-treated participants with Sjögren's Disease can:
 - Often maintain protective IgG levels to clinically relevant pathogens
 - Mount IgG responses to infection and vaccination
 - Follow recommended vaccination schedules

Background

- Nipocalimab is a fully human, high affinity, aglycosylated, effectorless, immunoselective immunoglobulin (Ig) G1 monoclonal antibody that blocks the neonatal fragment crystallizable receptor (FcRn), thereby lowering serum IgG levels[1-4]
- In the DAHLIAS study (NCT04968912), significant clinical improvement over placebo in Clinical European League Against Rheumatism Sjögren's Syndrome Disease Activity Index (ClinESSDAI) scores at 24 weeks were observed with nipocalimab 15 mg/kg intravenous (IV) biweekly treatment, which was well tolerated in participants with moderate-to-severe Sjögren's Disease (SjD)[5]
- Results from a randomized, open-label vaccine study in healthy volunteers suggest that nipocalimab does not impact the development of IgG responses to T-cell–dependent or –independent vaccines[6]

Objectives

To assess the impact of nipocalimab on pre-existing anti-vaccine antibodies to tetanus toxoid (TT) and varicella zoster virus (VZV), and the humoral responses to Severe Acute Respiratory Syndrome Coronavirus-2 (SARS-CoV-2) infection and vaccination in a post-hoc analysis of participants enrolled in the DAHLIAS study

Methods

Study Design

- In the Phase 2 DAHLIAS study (NCT04968912), participants with moderate-to-severe SjD received an IV injection of nipocalimab 5 mg/kg, 15 mg/kg, or placebo every 2 weeks for 22 weeks (**Figure 1**)
- Serum IgG antibody levels against TT and VZV were measured in baseline and post-treatment samples from a subset of participants who received 15 mg/kg nipocalimab
- In participants with documented SARS-CoV-2 vaccination or infection during the study, antibodies against different epitopes of SARS-CoV-2 were measured
- Participants in the placebo and the 15 mg/kg nipocalimab groups with available samples were included in the analysis

FIGURE 1: Study design

Key Eligibility Criteria

- ✓ Adults 18–75 years
- ✓ Anti-SSA positive, moderate-to-severe SjD
- ✓ Total ClinESSDAI score ≥6

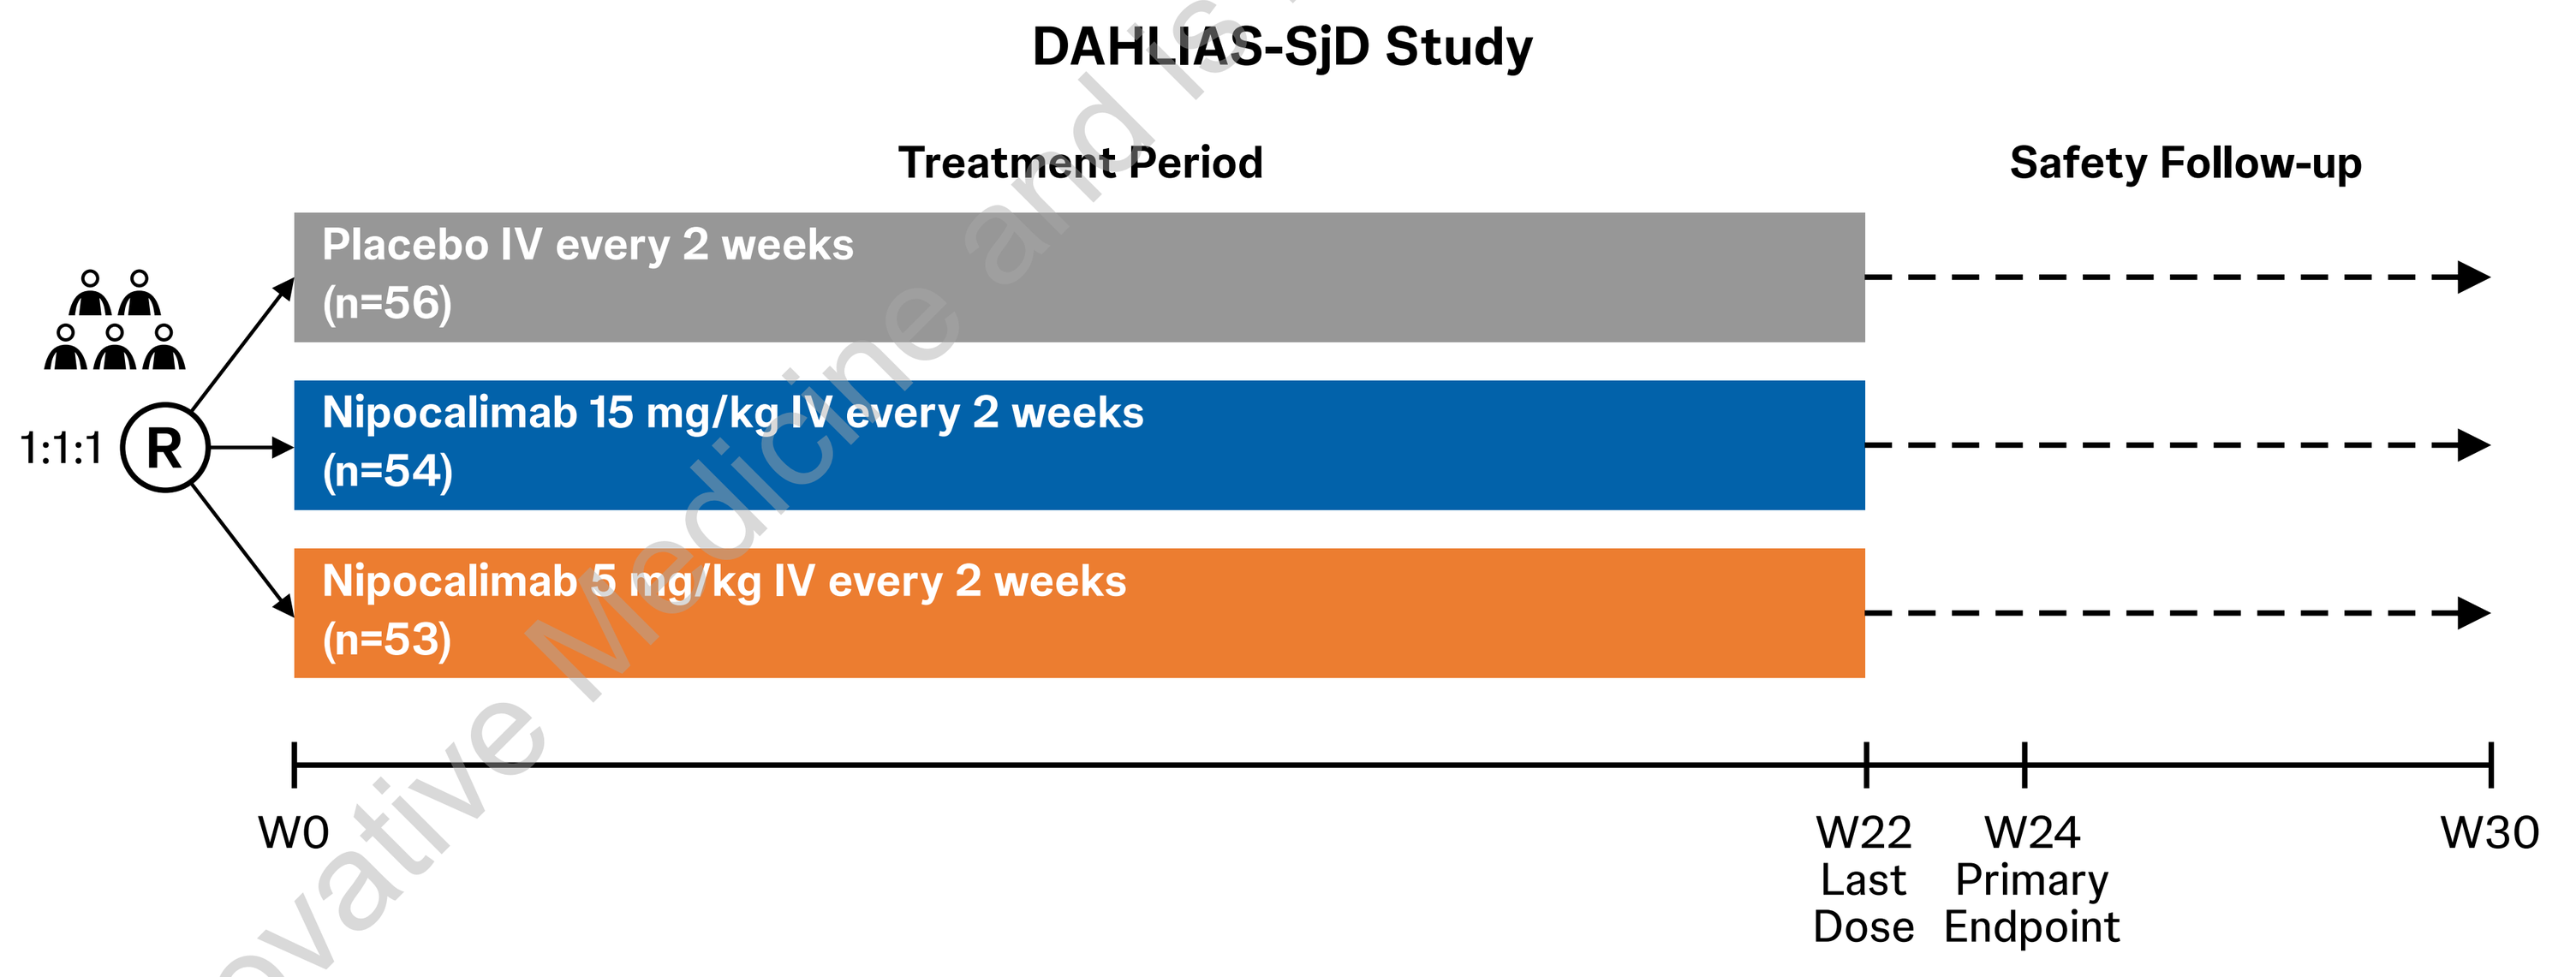
DAHLIAS-SjD Study
Treatment Period
Safety Follow-up
1:1:1
R
Placebo IV every 2 weeks (n=56)
Nipocalimab 15 mg/kg IV every 2 weeks (n=54)
Nipocalimab 5 mg/kg IV every 2 weeks (n=53)
W0
W22 Last Dose
W24 Primary Endpoint
W30

Anti-SSA=anti-Sjögren's syndrome-related antigen, ClinESSDAI=Clinical European League Against Rheumatism Sjögren's Syndrome Disease Activity Index, IV=intravenous, R=randomization, SjD=Sjögren's Disease, W=week.

Results

Nipocalimab treatment significantly and reversibly reduced total IgG levels in participants with SjD

- Nipocalimab 15 mg/kg produced a minimum reduction in total IgG of 61%, which was consistent with its mechanism of action (**Figure 2**)

FIGURE 2: Observed pre-dose (minimal) reduction in total IgG in participants with SjD

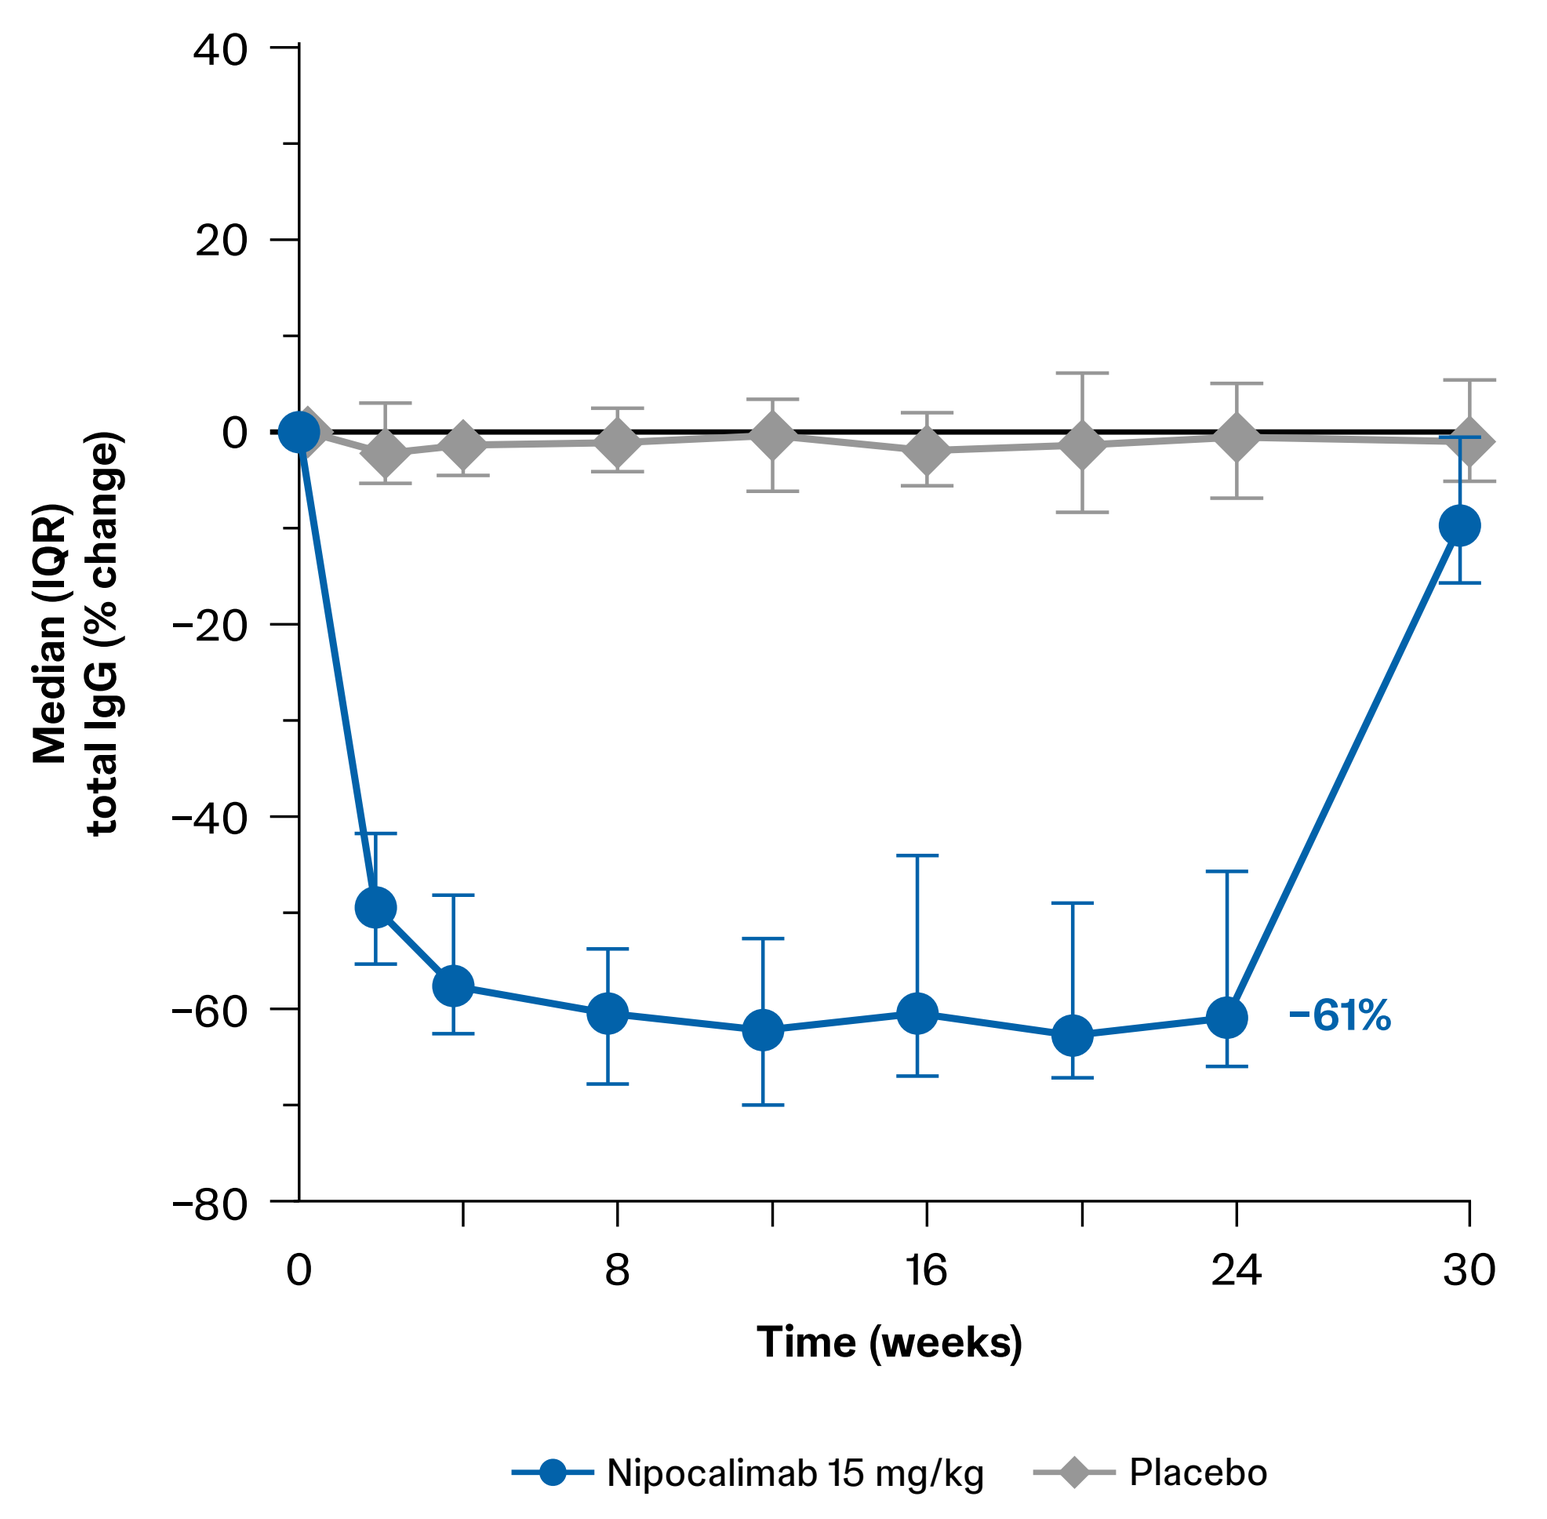
Median (IQR) total IgG (% change)
40
20
0
−20
−40
−60
−80
0
8
16
24
30
Time (weeks)
−61%
Nipocalimab 15 mg/kg
Placebo

IgG=immunoglobulin G, IQR=interquartile range, SjD=Sjögren's Disease.

Most participants remained protected during the study, despite reductions in pre-existing anti-TT IgG and anti-VZV IgG due to nipocalimab

- Nipocalimab reduced pre-existing anti-TT and anti-VZV consistent with the mechanism of action of nipocalimab in the DAHLIAS study (**Figure 3**)
- Nipocalimab reduced anti-TT IgG (**Figure 3A**) and anti-VZV IgG (**Figure 3C**) levels to a similar extent as total IgG levels; the majority of participants were able to maintain anti-TT IgG (**Figure 3B**) and anti-VZV IgG (**Figure 3D**) levels above the protective threshold
- Anti-TT IgG and anti-VZV IgG levels returned to baseline after treatment cessation (**Figures 3B and 3D**)

FIGURE 3: Levels of pre-existing anti-TT IgG (A and B) and anti-VZV (C and D) IgG during nipocalimab treatment

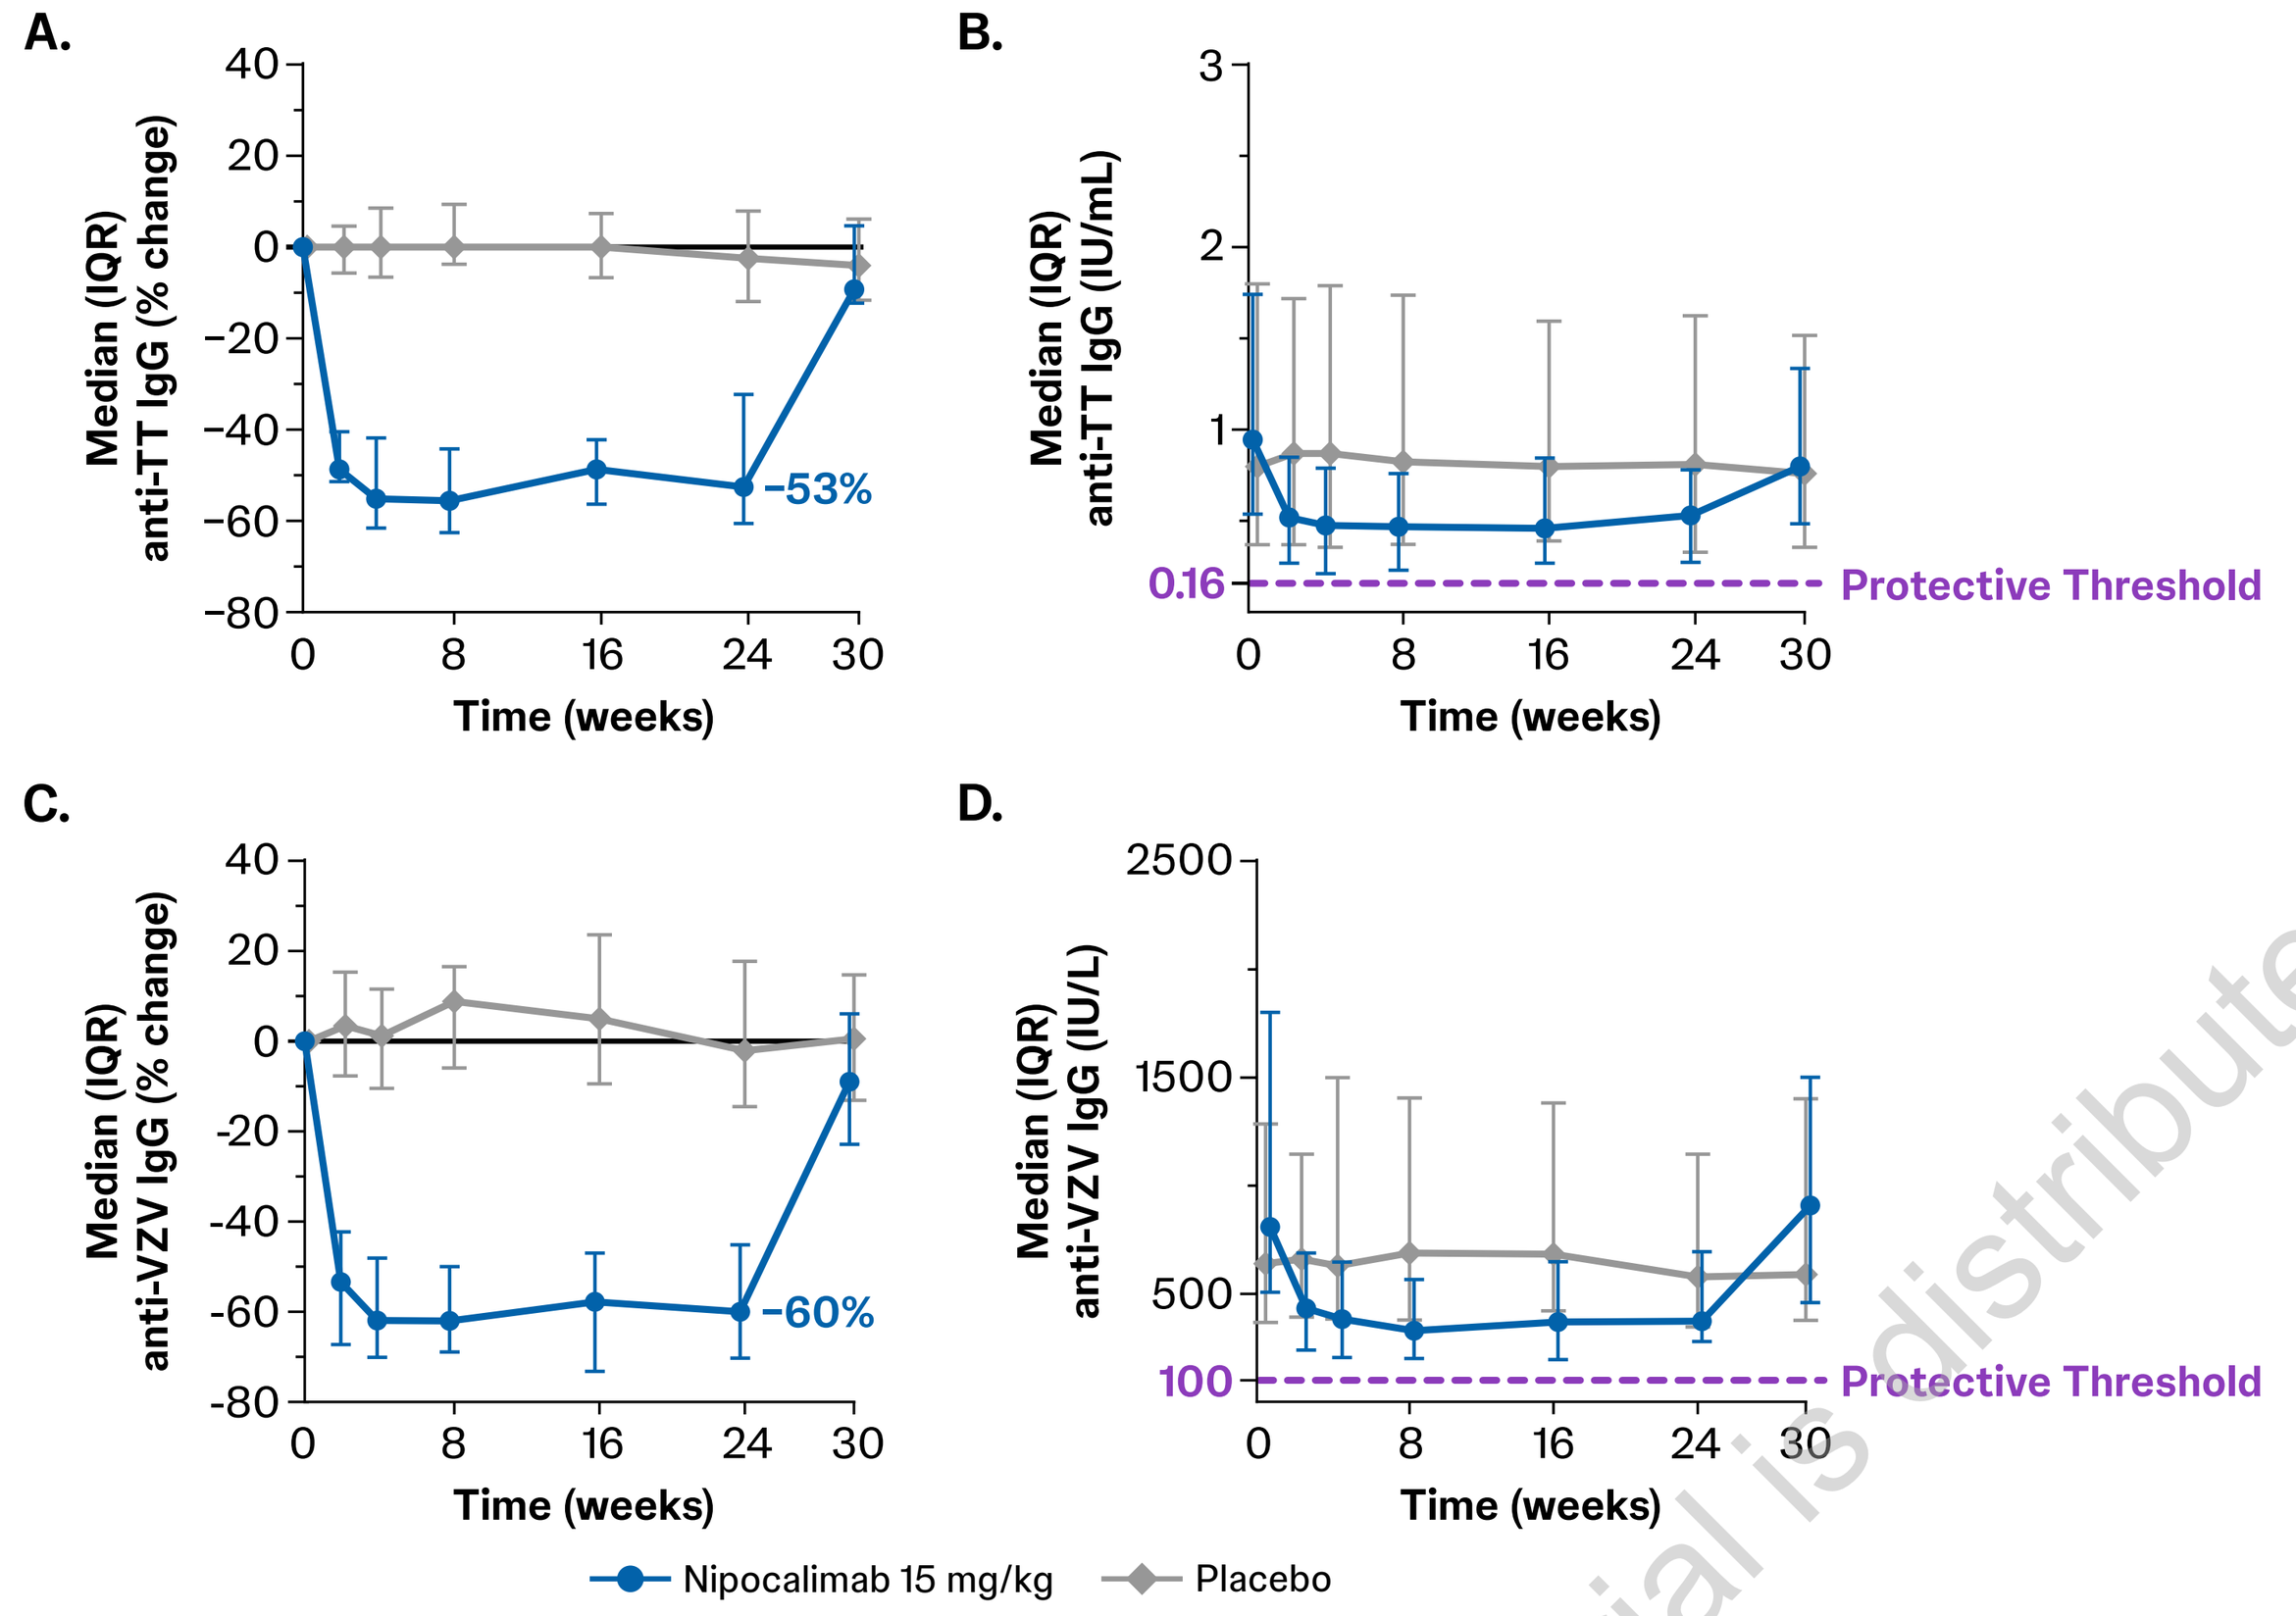
A.
Median (IQR) anti-TT IgG (% change)
−53%
B.
Median (IQR) anti-TT IgG (IU/mL)
0.16
Protective Threshold
C.
Median (IQR) anti-VZV IgG (% change)
−60%
D.
Median (IQR) anti-VZV IgG (IU/L)
100
Protective Threshold
Time (weeks)
Nipocalimab 15 mg/kg
Placebo

Treatment	Participants with anti-TT IgG levels above protective threshold (0.16 IU/mL), n/N (%)			
	BL/W0	W24	W30	All timepoints
Nipocalimab 15 mg/kg (n=41)	38	28/31 (90%)	30/31 (97%)	32/38 (84%)
Placebo (n=49)	44	38/39 (97%)	35/36 (97%)	42/44 (95%)

Treatment	Participants with anti-VZV IgG levels above protective threshold (100 IU/L), n/N (%)			
	BL/W0	W24	W30	All timepoints
Nipocalimab 15 mg/kg (n=50)	46	36/40 (90%)	39/39 (100%)	40/46 (87%)
Placebo (n=55)	53	46/46 (100%)	44/44 (100%)	53/53 (100%)

BL=baseline, IgG=immunoglobulin G, IQR=interquartile range, TT=tetanus toxoid, VZV=varicella zosters virus, W=week.

Nipocalimab-treated participants mounted IgG responses to SARS-CoV-2 antigens during acute infection and upon vaccination

- Participants with SARS-CoV-2 acute infection in the nipocalimab 15 mg/kg (n=3; 2 mild, 1 moderate) had increased levels of anti-spike and anti-nucleocapsid antibodies; all infections resolved without complications (**Figure 4A**)
- All participants receiving SARS-CoV-2 vaccination mounted a humoral response as shown by increases in anti-spike antibodies; no humoral responses were observed against nucleocapsid, since nucleocapsid is not part of the vaccine (**Figure 4B**)

FIGURE 4: Levels of anti-spike/S1 RBD/nucleocapsid IgG and total IgG during nipocalimab treatment following COVID-19 infection (A) and COVID-19 vaccination (B)

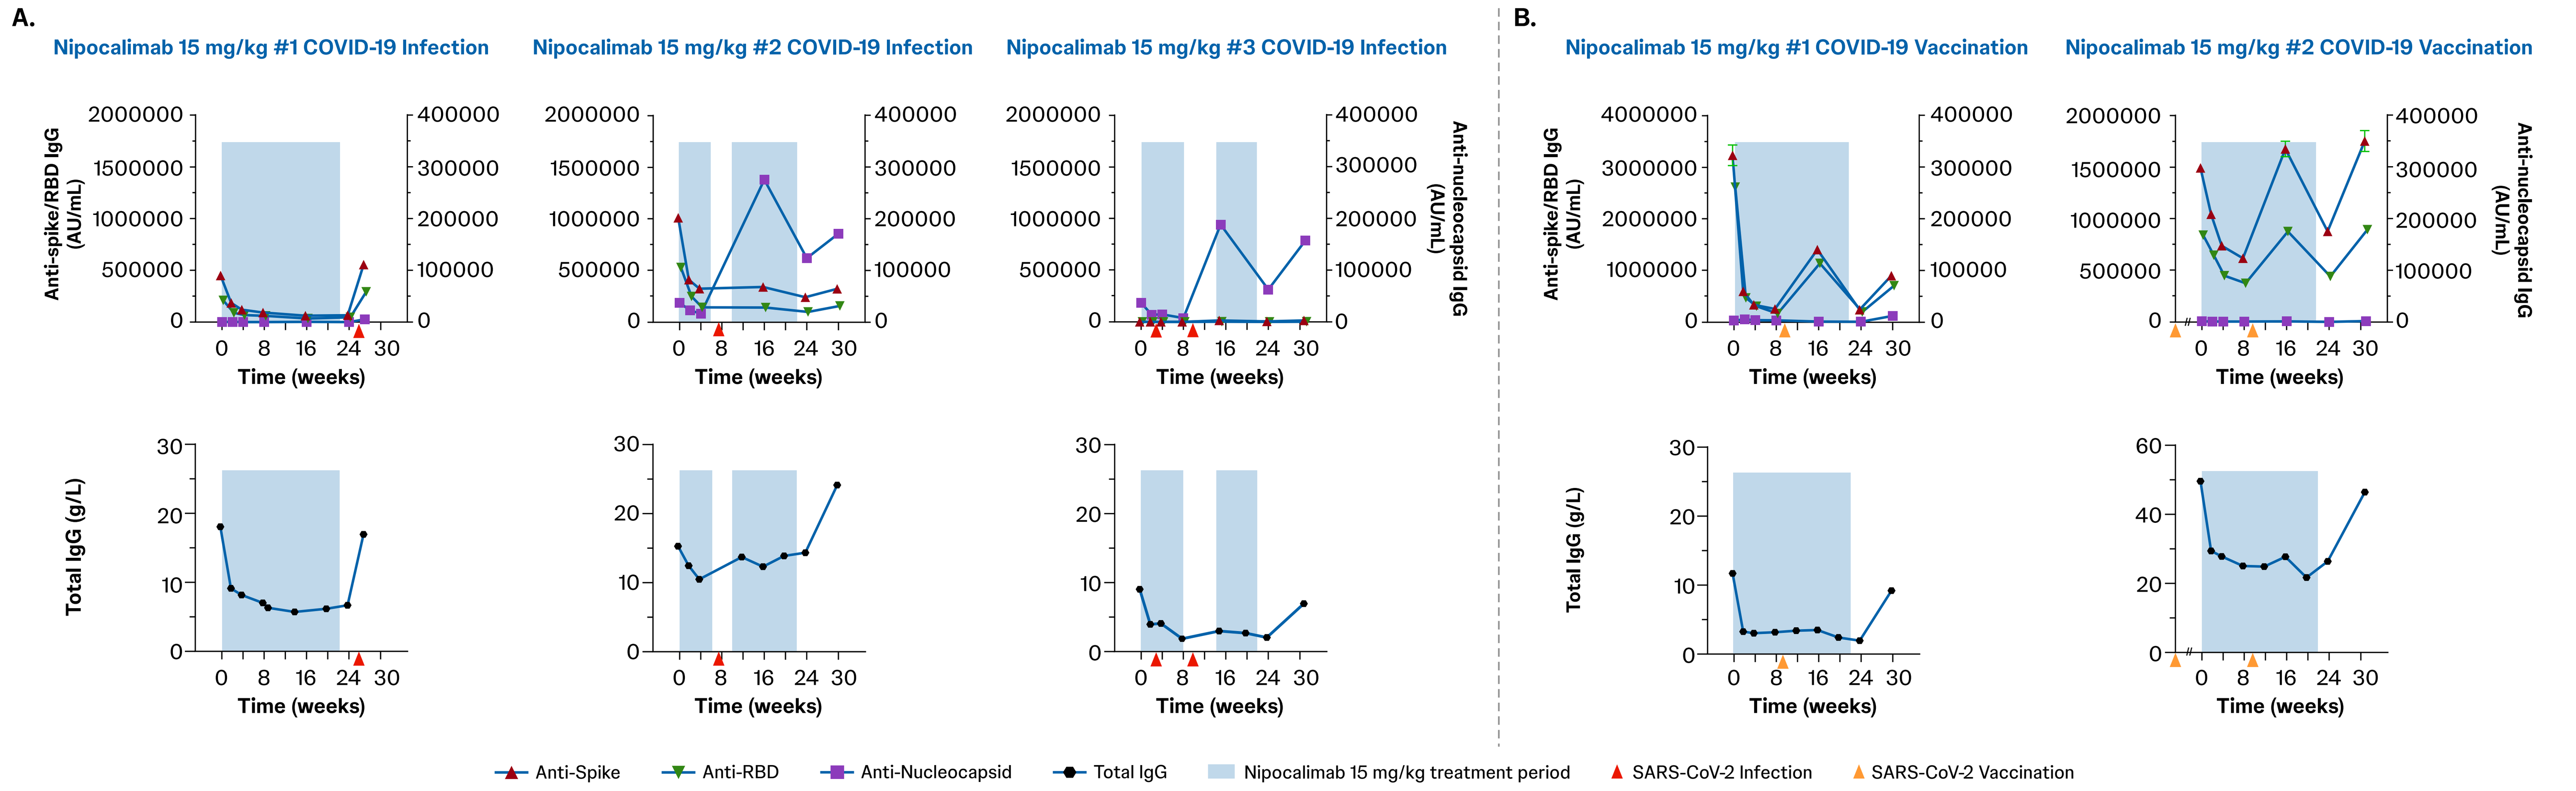
A.
Nipocalimab 15 mg/kg #1 COVID-19 Infection
Nipocalimab 15 mg/kg #2 COVID-19 Infection
Nipocalimab 15 mg/kg #3 COVID-19 Infection
B.
Nipocalimab 15 mg/kg #1 COVID-19 Vaccination
Nipocalimab 15 mg/kg #2 COVID-19 Vaccination
Anti-spike/RBD IgG (AU/mL)
Anti-nucleocapsid IgG (AU/mL)
Total IgG (g/L)
Time (weeks)
Anti-Spike
Anti-RBD
Anti-Nucleocapsid
Total IgG
Nipocalimab 15 mg/kg treatment period
SARS-CoV-2 Infection
SARS-CoV-2 Vaccination

AU=Arbitrary unit, COVID-19=Coronavirus disease 2019, IgG=immunoglobulin G, RBD=receptor-binding domain, SARS-CoV-2=Severe acute respiratory syndrome coronavirus 2.

PRESENTED BY: Kathleen A. Packard at the Congress of Clinical Rheumatology-West (CCR-West), September 17–20, 2026; Huntington Beach, California, USA. **REFERENCES: 1.** Ling LE, et al. *Clin Pharmacol Ther.* 2019;105(4):1031–1039. **2.** Roy S, et al. *Am J Obstet Gynecol.* 2019;220(5):498e.1–498.e9. **3.** Moise KJ Jr, et al. *N Engl J Med.* 2024;391(6):526–537. **4.** Antozzi C, et al. *Neurology.* 2024;102(2):e207937. **5.** Cossu M, et al. *Hum. Vaccin. Immunother.* 2025;21(1):2491269. **6.** Gottenberg J, et al. *Arthritis Rheumatol.* 2024; 76(suppl 9). **ACKNOWLEDGMENTS:** Medical writing support was provided by Jennifer L Venzie, PhD (SystemOne) under the direction of the authors in accordance with Good Publication Practice guidelines (*Ann Intern Med.* 2022;175:1298–1304). Layout design and formatting for this presentation were provided by Sandeep Chavan of SIRO Medical Writing Pvt. Ltd., India. This study and presentation were sponsored by Johnson & Johnson. **DISCLOSURES: FY, EM, JJH, KC, MJL, KAP, DD, CC,** and **SG** are employees of Johnson & Johnson and may hold stock in Johnson & Johnson. **PREVIOUSLY PRESENTED AT:** American College of Rheumatology (ACR) Convergence; October 24–29, 2025; Chicago, IL, USA.