Biomarker-Driven Insights to Clinical Response in DAHLIAS: A Nipocalimab Trial for Sjögren's Disease

Hal Scofield,[1,2] He Li,[3] Jacques-Eric Gottenberg,[4] David Galbraith,[3] Keying Ma,[3] Lynn Tomsho,[3] Chris Sisk,[3] Kathleen A. Packard,[3] Matthew Loza,[3] Jonathan Hubbard,[3] Kim Campbell,[3] Dawn Waterworth,[3] Sheng Gao,[3] Carolyn Cuff,[5] Steven Leonardo,[5] Kathy Sivils[3]

[1]Arthritis & Clinical Immunology Program, Oklahoma Medical Research Foundation, Oklahoma City, OK, USA; [2]Department of Medicine, University of Oklahoma Health Sciences Center, Oklahoma City, OK, USA; [3]Johnson & Johnson, Spring House, PA, USA; [4]Strasbourg University Hospital, Strasbourg, France; [5]Johnson & Johnson, Cambridge, MA, USA

Scan the QR code.
The QR code is intended to provide scientific information for individual reference, and the information should not be altered or reproduced in any way.

This material is distributed for scientific purposes by Johnson & Johnson Innovative Medicine and is not for promotional use.

Key Takeaways

- **In DAHLIAS, nipocalimab 15 mg/kg IV Q2W reduced ClinESSDAI score and total IgG significantly in participants with SjD, with a safety profile consistent with other phase 2 studies[8–10]**
- **Biomarker analyses reinforce the mechanistic rationale for treatment with nipocalimab in SjD, with enriched response observed among participants with elevated aAbs, IFN, and IgG levels**
- **This supports the continued development of nipocalimab in SjD in the ongoing phase 3 DAFFODIL study (NCT06741969)**

Background

- Sjögren's Disease (SjD) is characterized by presence of autoantibody (aAb), lymphocytic infiltration of exocrine glandular tissues, immune complex formation and interferon (IFN) production[1]
- Systemic immune dysregulation involves aberrant B-lymphocyte activity and abnormally elevated immunoglobulin G (IgG) and IgG aAb levels (eg, anti-Ro, anti-La)[1]
- SjD is associated with a relentless multi-organ burden, with ≥1 organ involvement in approximately 40% to 75% of patients, commonly manifested by mucosal dryness, fatigue, and pain, and an approximately 1.5-fold increase in all-cause mortality[2–7]

Objective

To evaluate whether nipocalimab's mechanism of action and SjD pathology impact response to nipocalimab in participants with SjD in the phase 2 DAHLIAS study, using biomarker stratification approaches

Methods

Study population
- Adults aged 18–75 years
- Moderately to severely active primary SjD (total ClinESSDAI score ≥6)
- Seropositive for anti-Ro60 and/or anti-Ro52 IgG antibodies

Primary endpoint[a]
- Change from baseline in ClinESSDAI score at Week 24

Key secondary endpoints
- ClinESSDAI-4 response[b] at Week 24

Post hoc analyses in biomarker-based populations
- ClinESSDAI-4 response[b] at Week 24

DAHLIAS Study Design

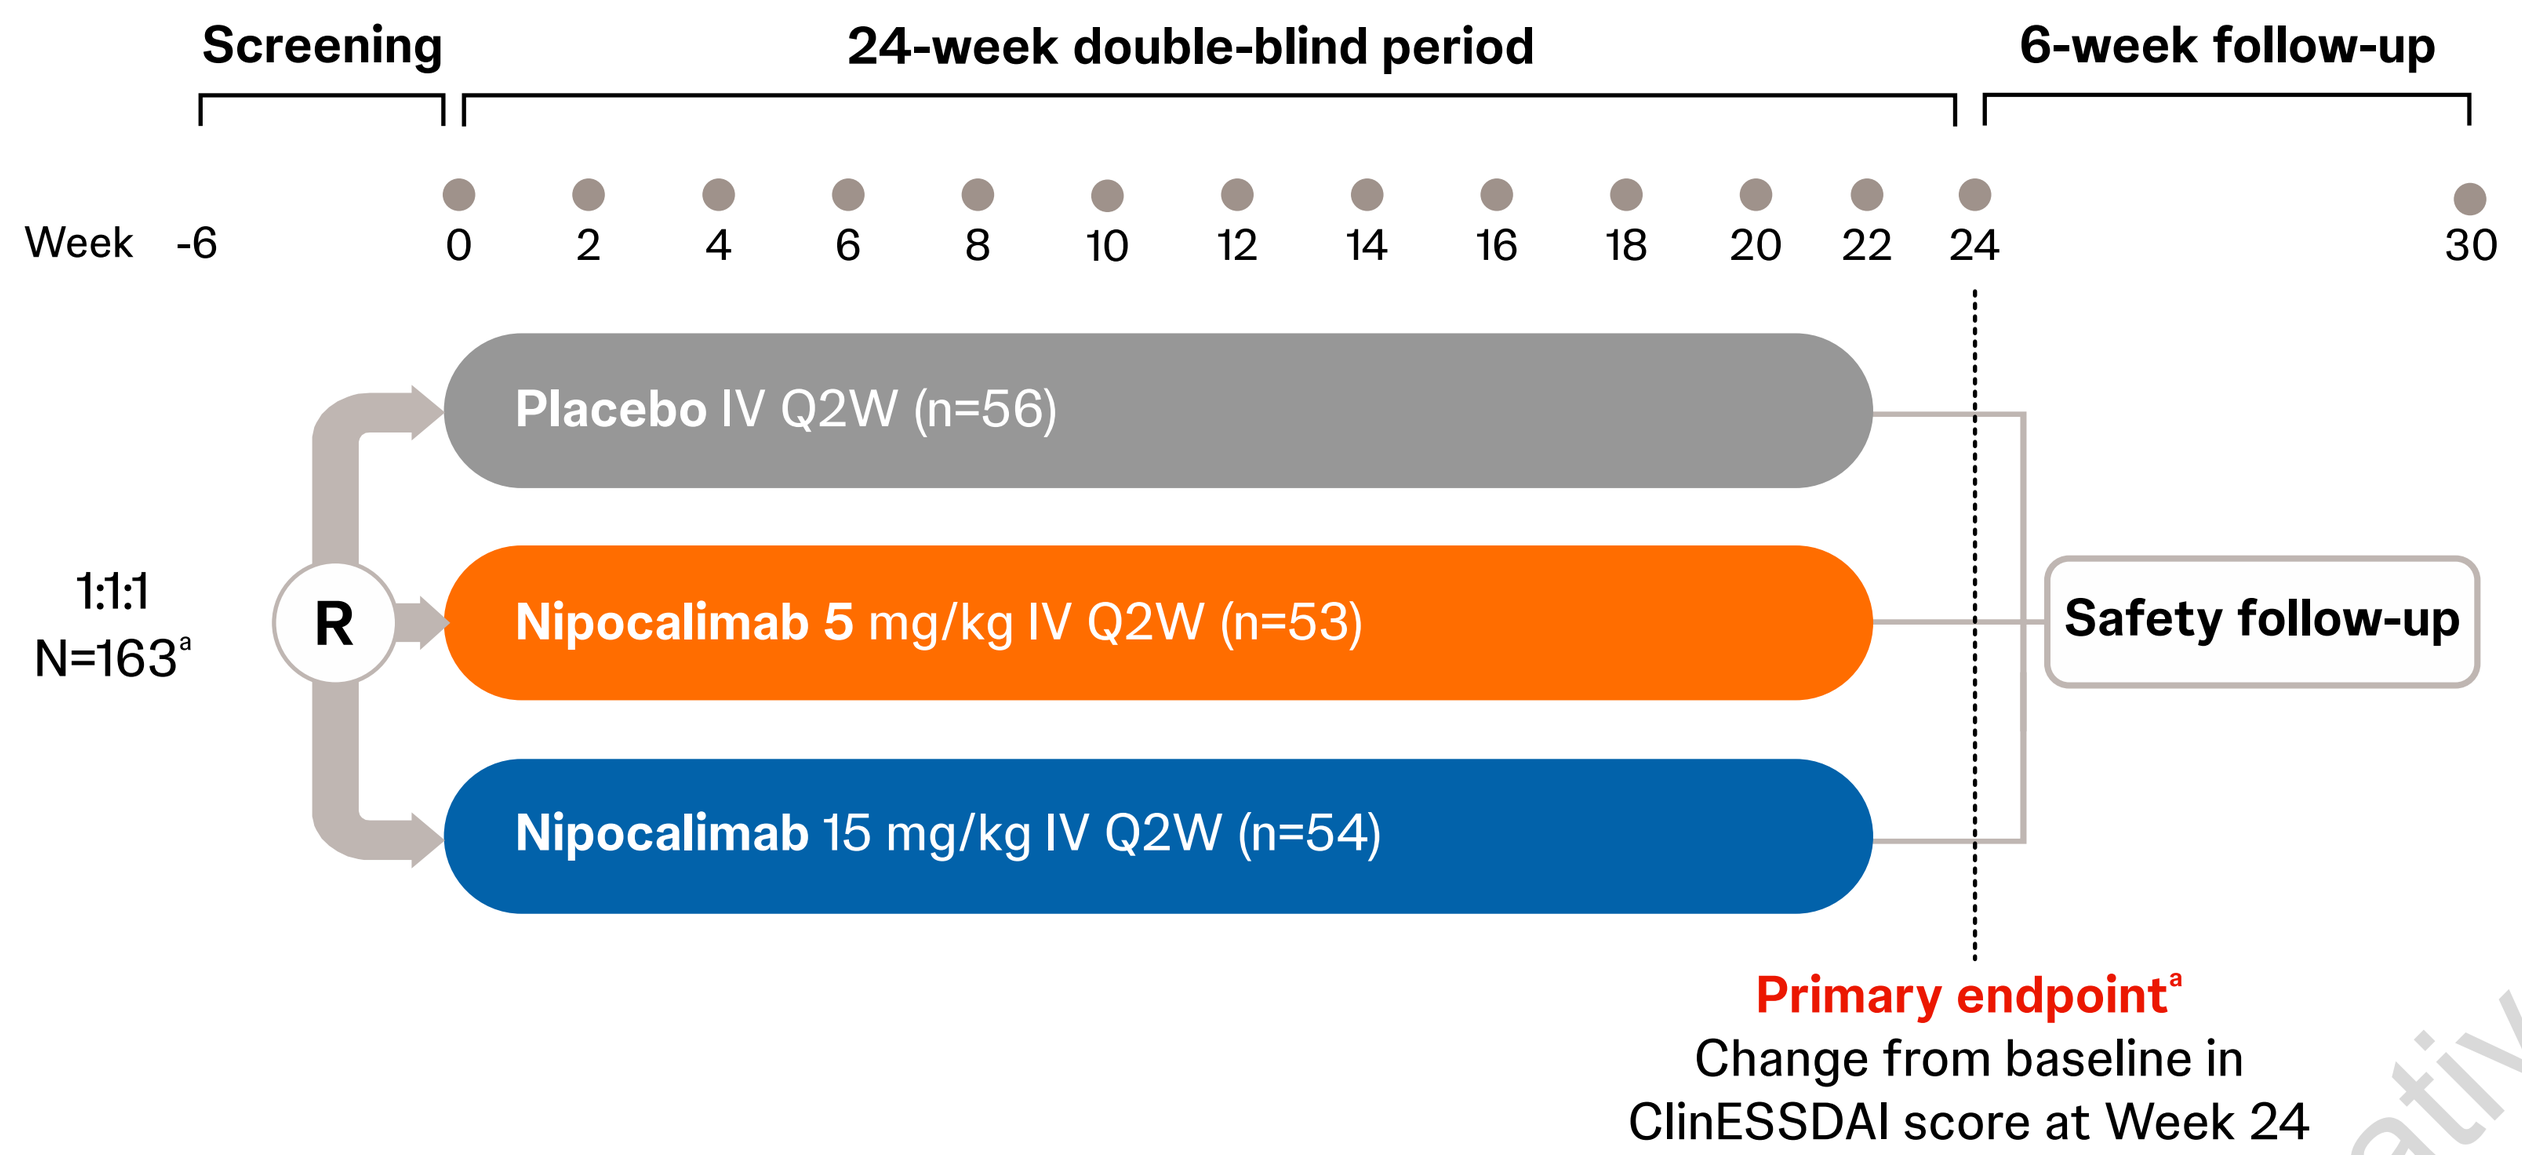
Screening
24-week double-blind period
6-week follow-up
Week -6 0 2 4 6 8 10 12 14 16 18 20 22 24 30
1:1:1
N=163[a]
R
Placebo IV Q2W (n=56)
Nipocalimab 5 mg/kg IV Q2W (n=53)
Nipocalimab 15 mg/kg IV Q2W (n=54)
Safety follow-up
Primary endpoint[a]
Change from baseline in ClinESSDAI score at Week 24

*[a]The DAHLIAS study was pre-specified to use a 2-sided alpha level of 0.1 without multiplicity control. [b]Defined as improvement of ≥4 points from baseline in ClinESSDAI score. **ClinESSDAI**=Clinical European Alliance of Associations for Rheumatology Sjögren's Syndrome Disease Activity Index, **IgG**=immunoglobulin G, **IV**=intravenous, **Q2W**=every 2 weeks, **R**=randomized, **SjD**=Sjögren's disease.*

Participant Stratification

DAHLIAS participants were stratified using 3 approaches based on nipocalimab's MOA and SjD pathology to test their impacts on response to nipocalimab:

Baseline aAbs
Serum baseline levels of anti-Ro60, anti-Ro52, and anti-La aAbs:
- **aAb-high:** all 3 aAb levels in the top tertile

Baseline IFN scores
IFN gene signature scores derived from whole-blood RNA sequencing:
- **IFN-high:** scores >75th percentile + 1.5 × IQR of healthy controls

Baseline IgG
Total serum IgG levels at baseline:
- **IgG-high:** IgG >25% quartile of overall study population

aAb=autoantibody, IFN=interferon, IgG=immunoglobulin G, IQR=interquartile range, MOA=mechanism of action, SjD=Sjögren's disease.

Results

DAHLIAS Primary Results: Efficacy and Safety

- **Safety:** Nipocalimab was well tolerated in participants with SjD, with no new safety signals observed

Primary endpoint:
Change from baseline in ClinESSDAI score at Week 24

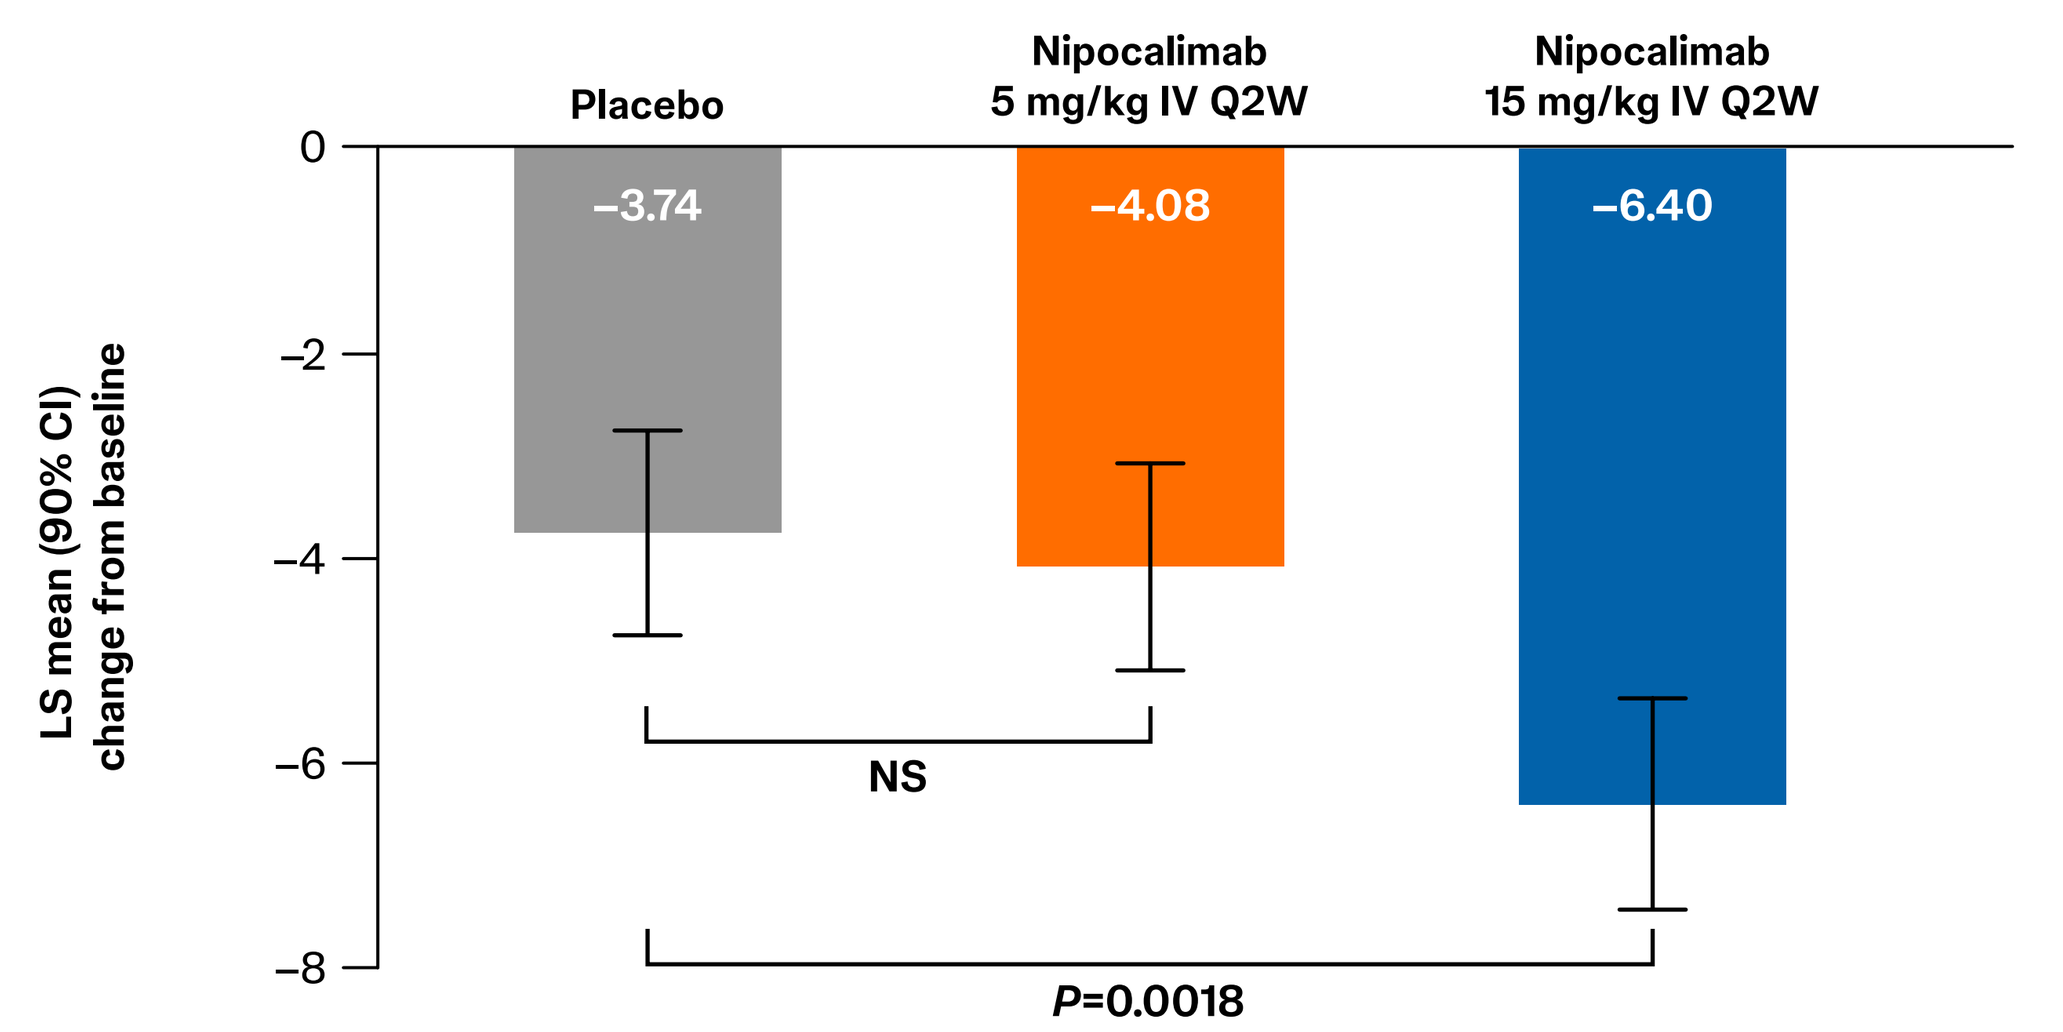
Placebo
Nipocalimab 5 mg/kg IV Q2W
Nipocalimab 15 mg/kg IV Q2W
−3.74
−4.08
−6.40
LS mean (90% CI) change from baseline
NS
P=0.0018

Secondary endpoint:
ClinESSDAI-4 response at Week 24

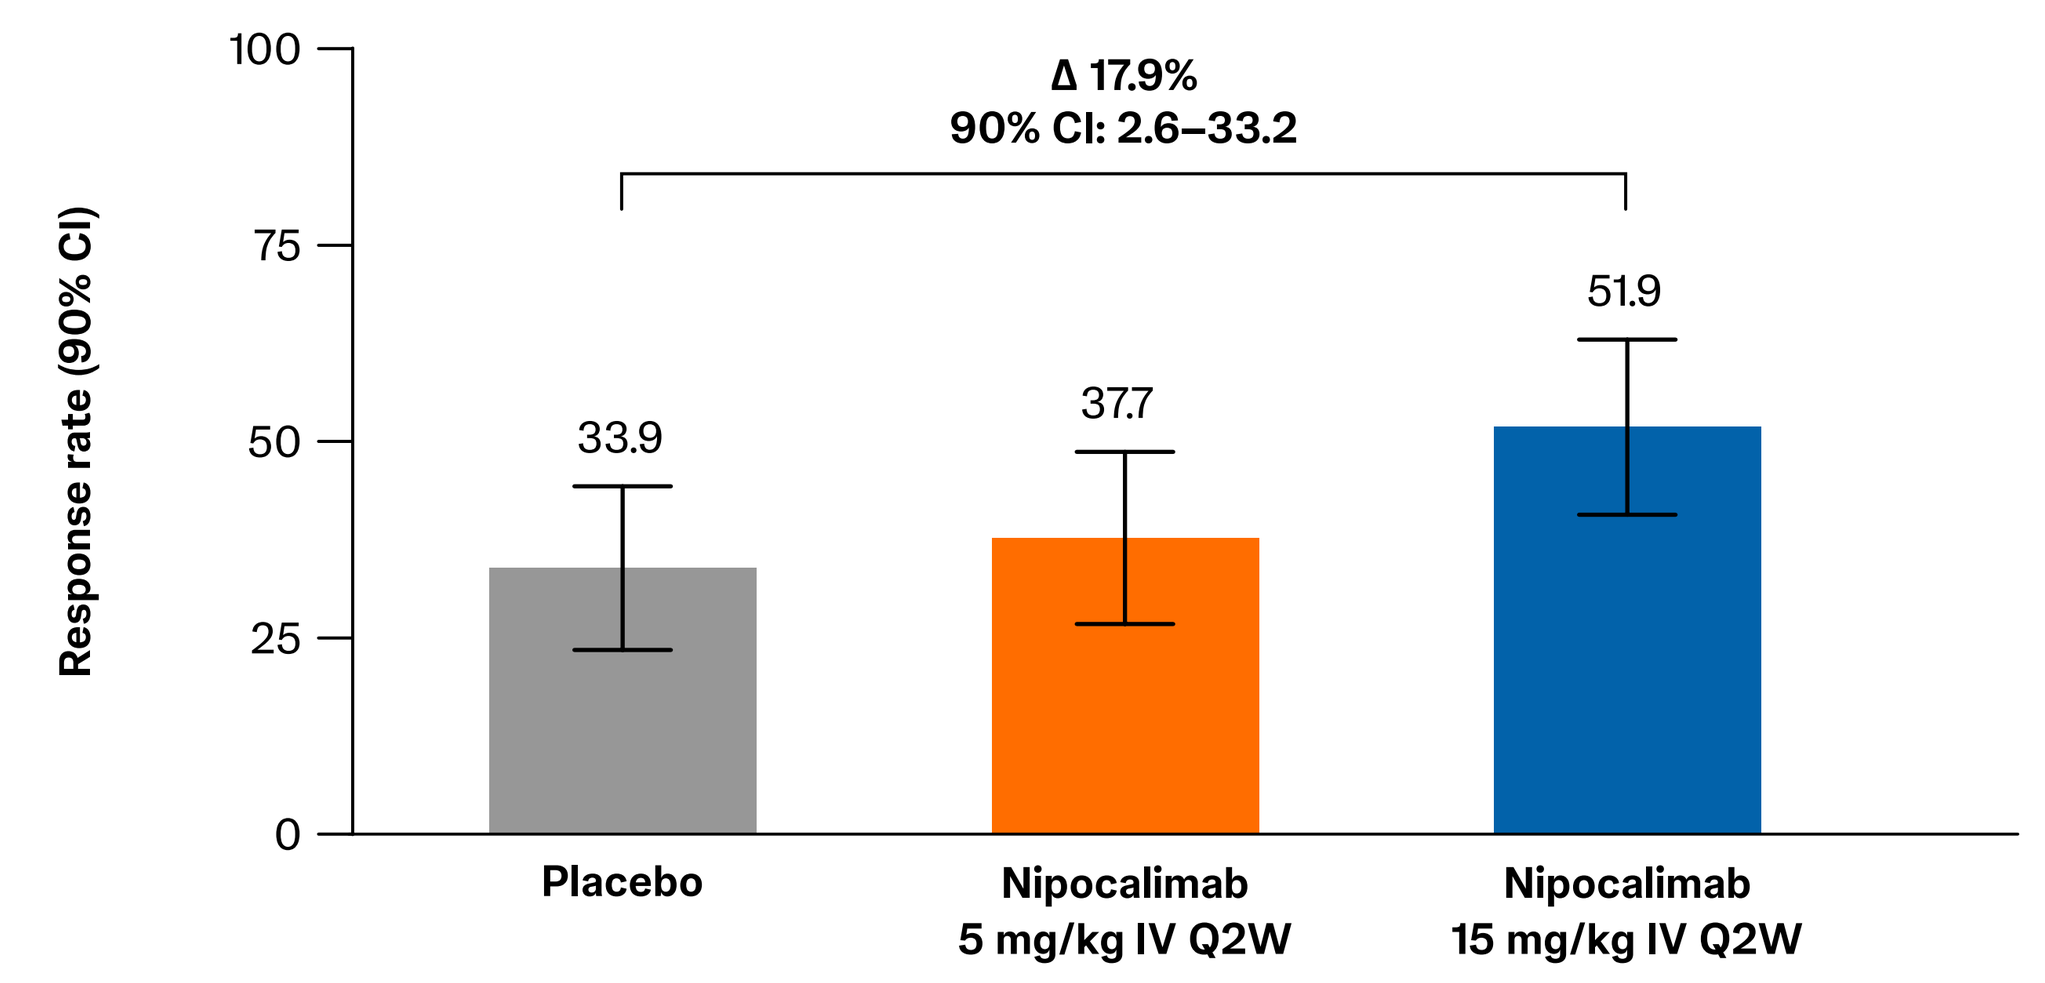
Δ 17.9%
90% CI: 2.6–33.2
33.9
37.7
51.9
Response rate (90% CI)
Placebo
Nipocalimab 5 mg/kg IV Q2W
Nipocalimab 15 mg/kg IV Q2W

CI=confidence interval, ClinESSDAI=Clinical European Alliance of Associations for Rheumatology Sjögren's Syndrome Disease Activity Index, IV=intravenous, LS=least squares, NS=nonsignificant, Q2W=every 2 weeks, SjD=Sjögren's disease.

DAHLIAS Primary Results: Biomarkers

Total IgG reduction
- Observed predose (minimum) median IgG reduction of 61% at Week 24
- 77% maximum reduction in total IgG (pharmacokinetic/pharmacodynamic [PK/PD] simulations)

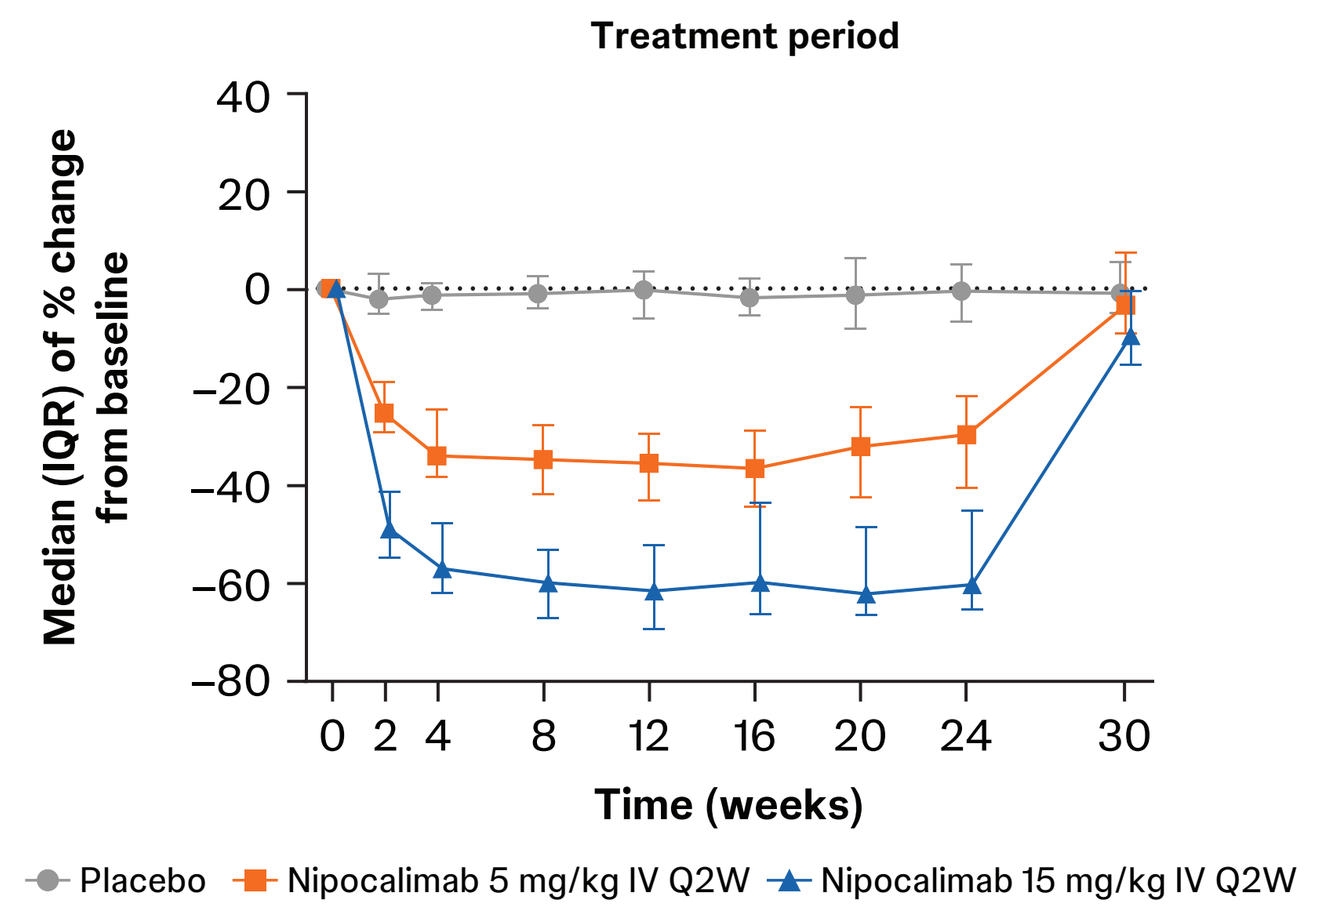
Treatment period
Median (IQR) of % change from baseline
Time (weeks)
Placebo
Nipocalimab 5 mg/kg IV Q2W
Nipocalimab 15 mg/kg IV Q2W

Anti-Ro60 IgG reduction
- Observed consistent reductions in SjD-associated anti-Ro60, anti-Ro52, and anti-La IgG aAbs during treatment

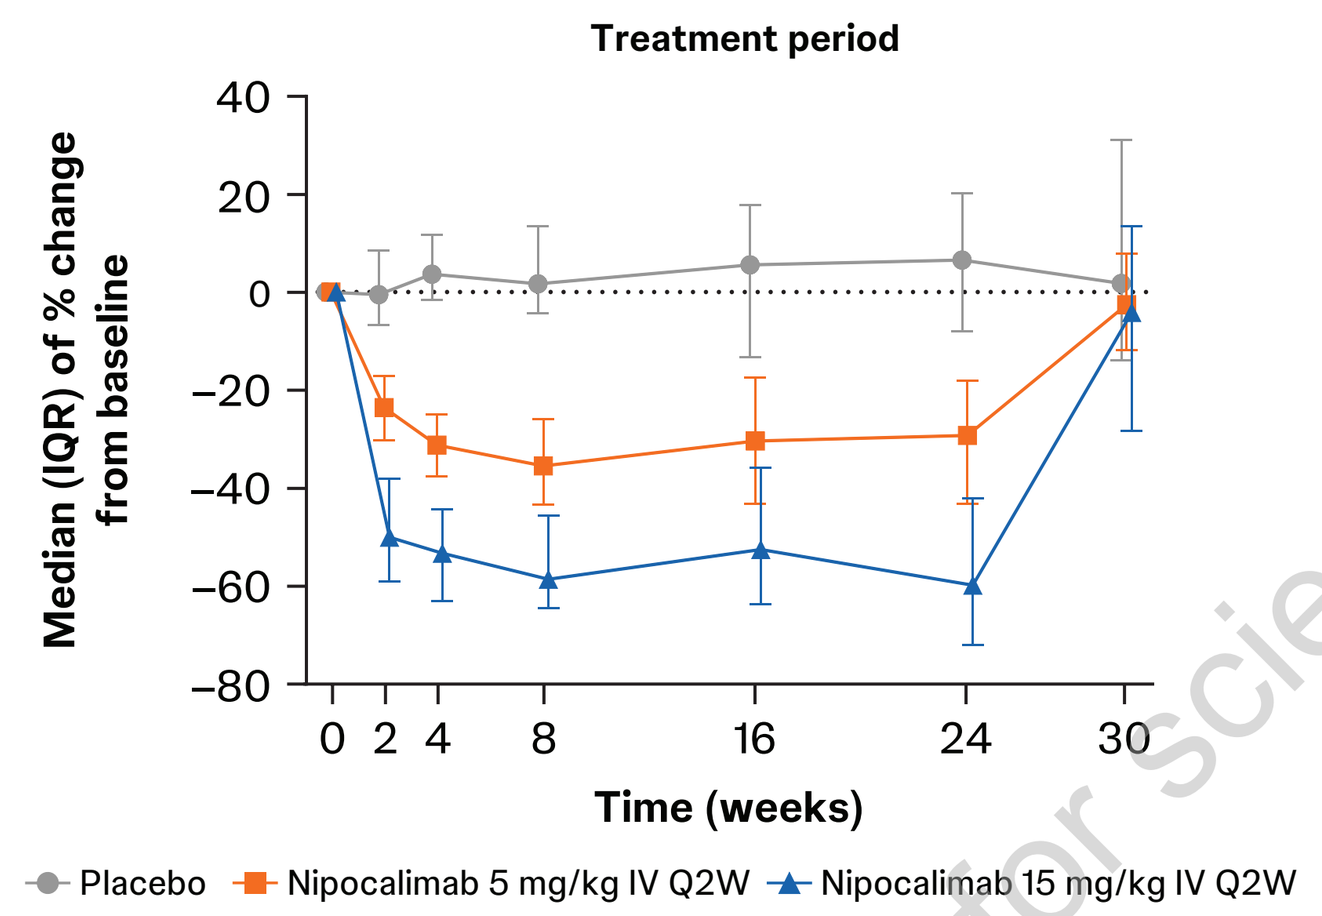
Treatment period
Median (IQR) of % change from baseline
Time (weeks)
Placebo
Nipocalimab 5 mg/kg IV Q2W
Nipocalimab 15 mg/kg IV Q2W

CIC (circulating IgG immune complexes) reduction
- Observed reductions in CICs during treatment

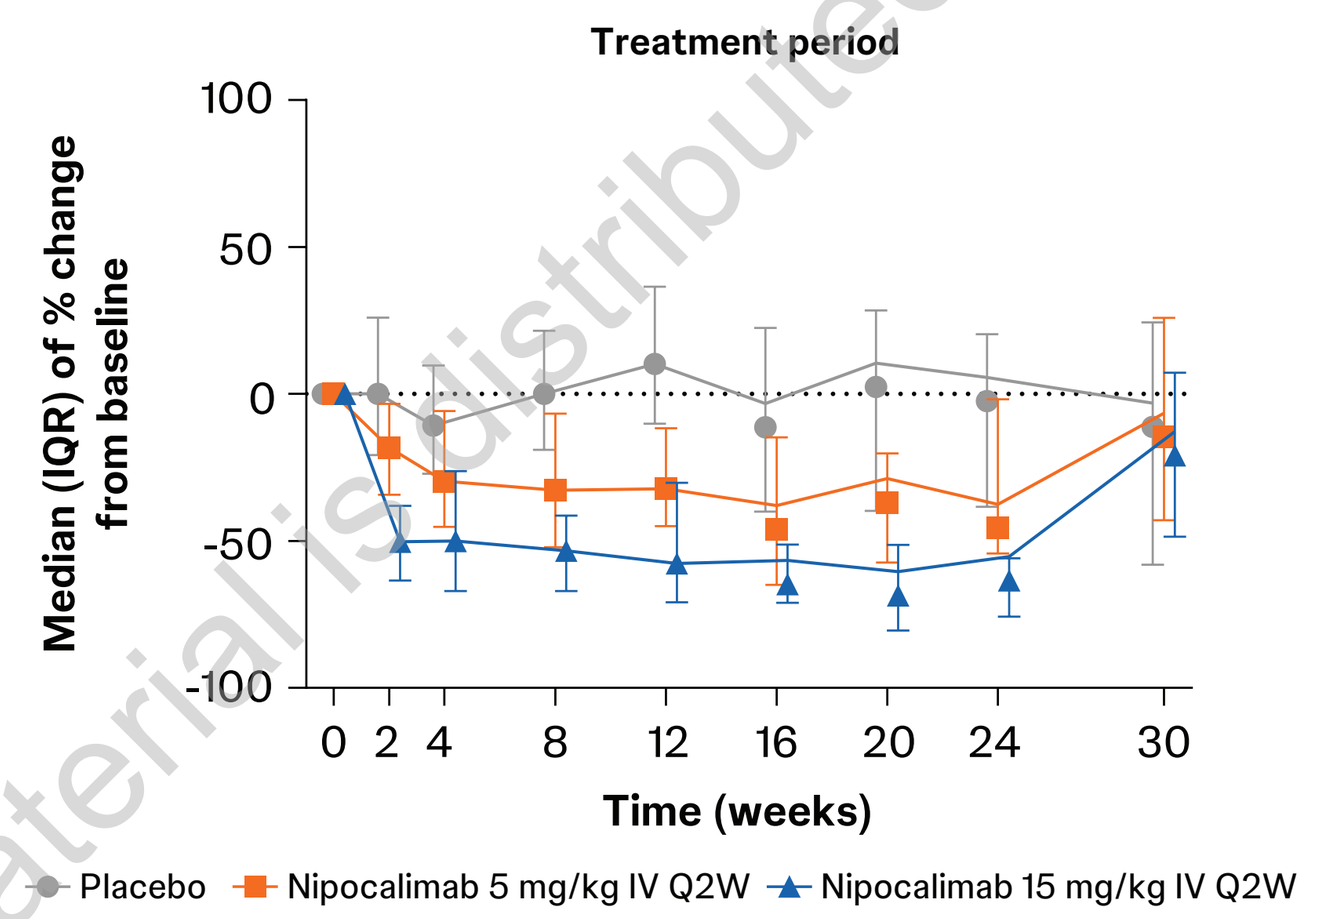
Treatment period
Median (IQR) of % change from baseline
Time (weeks)
Placebo
Nipocalimab 5 mg/kg IV Q2W
Nipocalimab 15 mg/kg IV Q2W

aAb=autoantibody, CIC=circulating IgG immune complexes, IgG=immunoglobulin G, IQR=interquartile range, IV=intravenous, Q2W=every 2 weeks, SjD=Sjögren's disease.

High Baseline aAb Level Enriches Responders to Nipocalimab

- Enriched nipocalimab response in aAb-high population (15% of DAHLIAS participants)

Baseline aAb levels

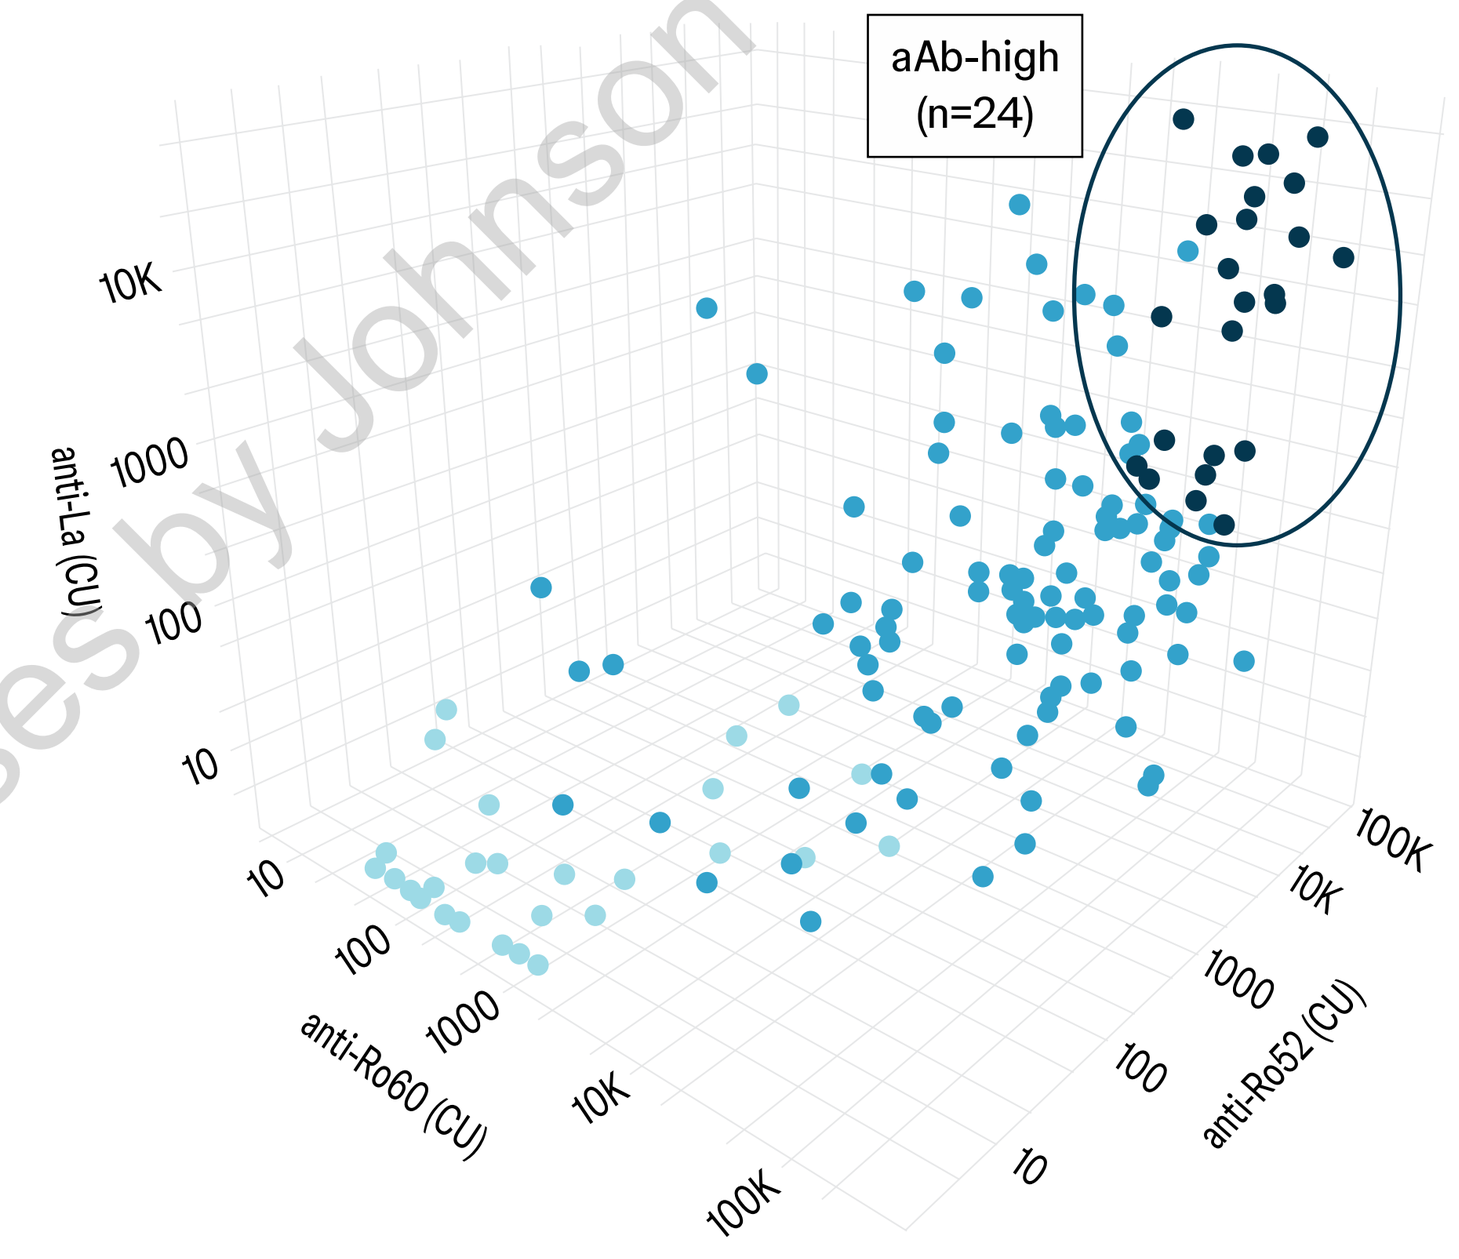
aAb-high
(n=24)
anti-La (CU)
anti-Ro60 (CU)
anti-Ro52 (CU)

ClinESSDAI-4 responses at Week 24 in aAb-high participants

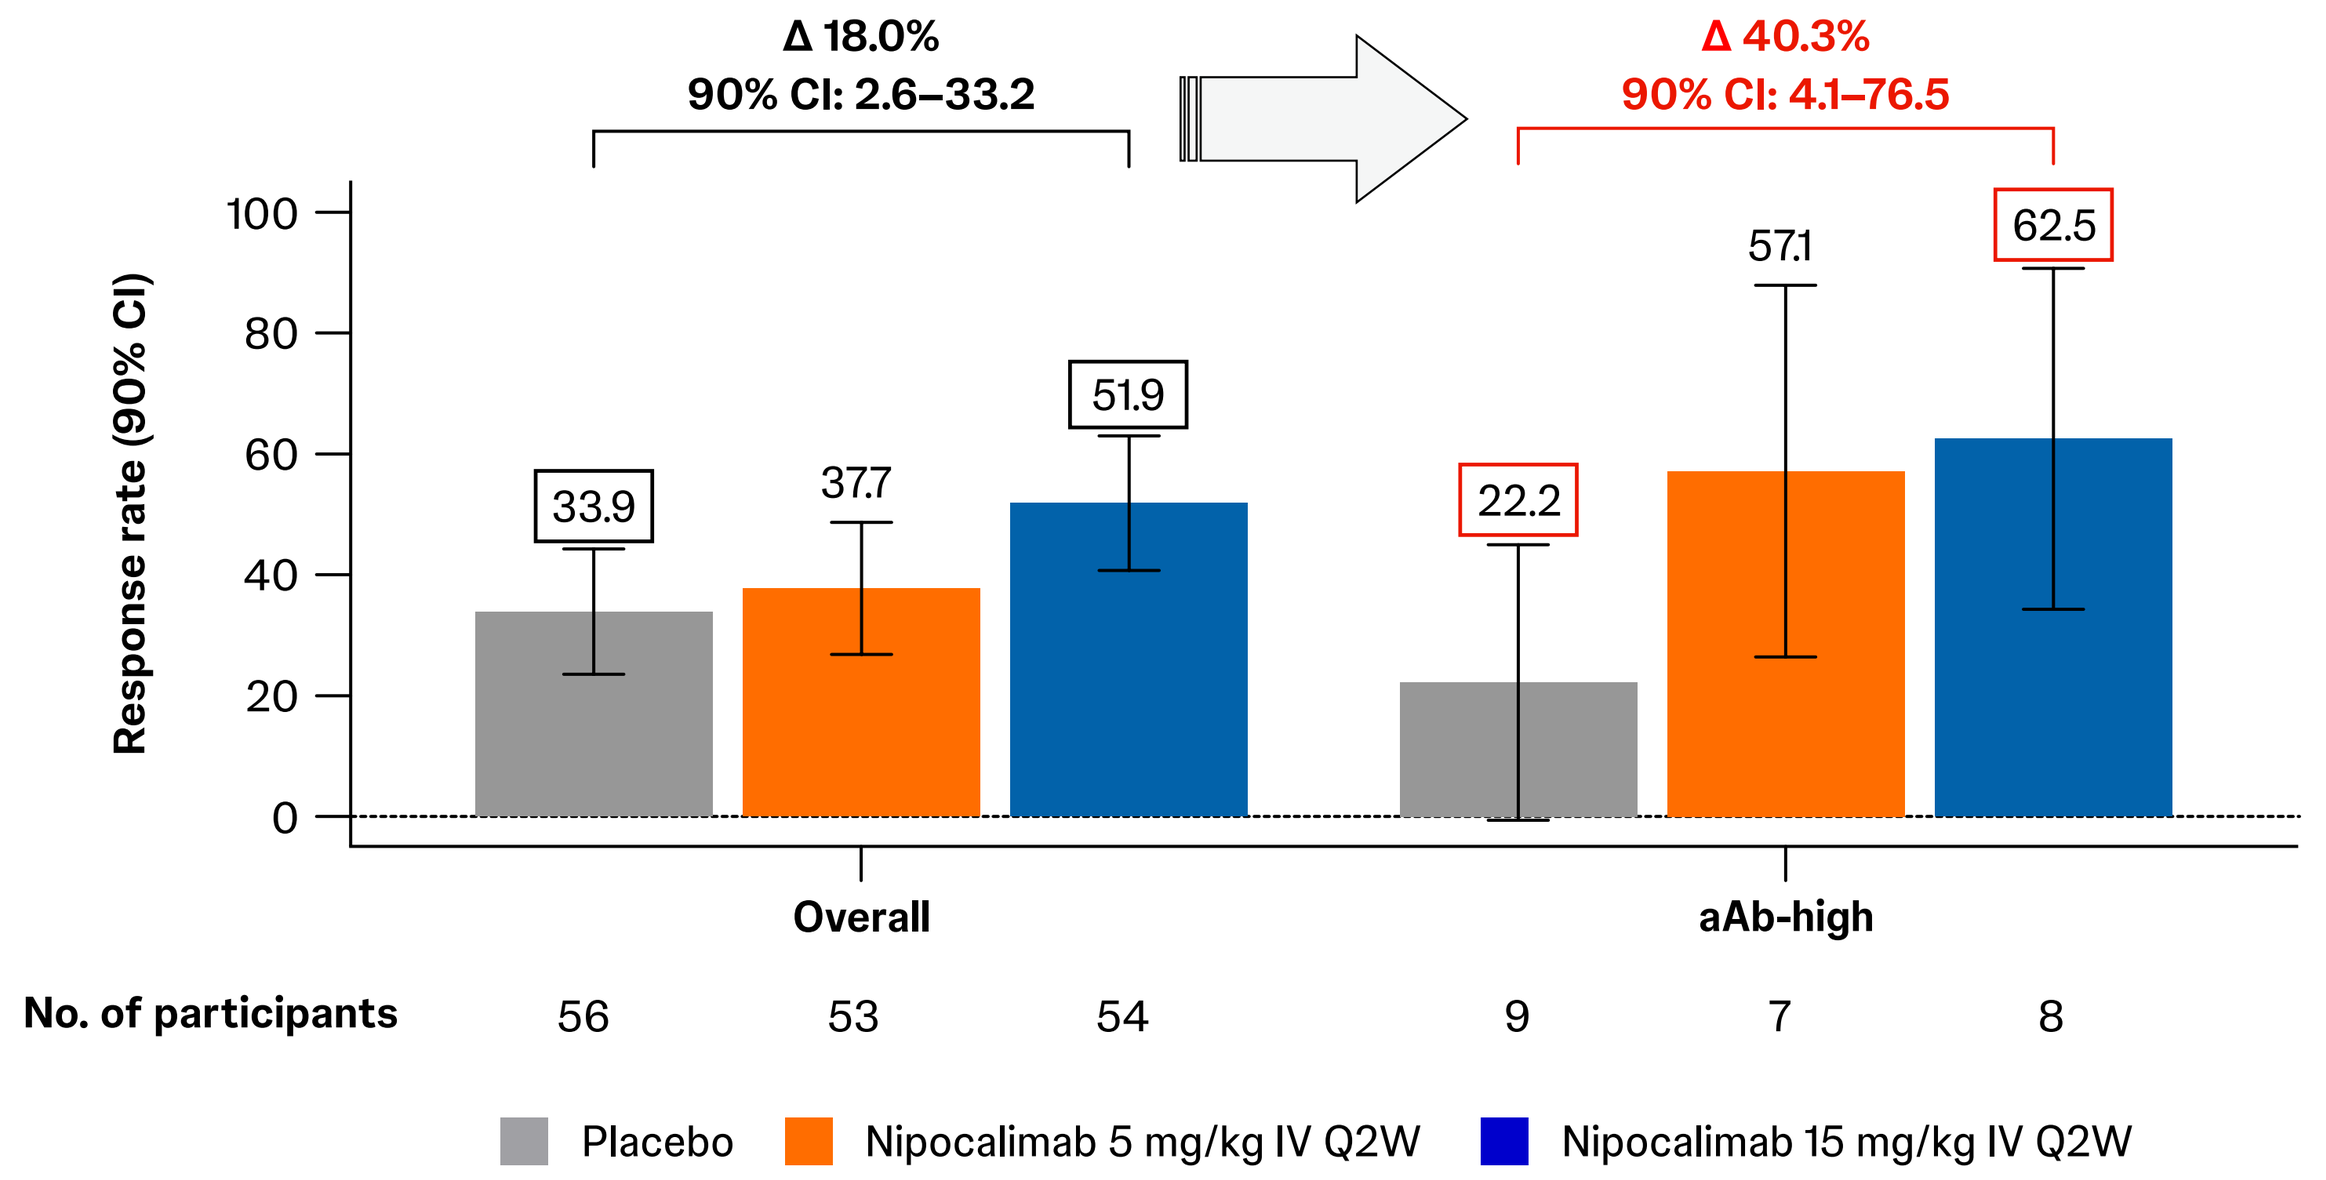
Δ 18.0%
90% CI: 2.6–33.2
Δ 40.3%
90% CI: 4.1–76.5
Response rate (90% CI)
33.9
37.7
51.9
22.2
57.1
62.5
Overall
aAb-high
No. of participants 56 53 54 9 7 8
Placebo
Nipocalimab 5 mg/kg IV Q2W
Nipocalimab 15 mg/kg IV Q2W

aAb=autoantibody, CI=confidence interval, ClinESSDAI=Clinical European Alliance of Associations for Rheumatology Sjögren's Syndrome Disease Activity Index, CU=chemiluminescent unit, IV=intravenous, Q2W=every 2 weeks.

High Baseline IFN Score Enriches Responders to Nipocalimab

- Enriched nipocalimab response in IFN-high population (74% of DAHLIAS participants)

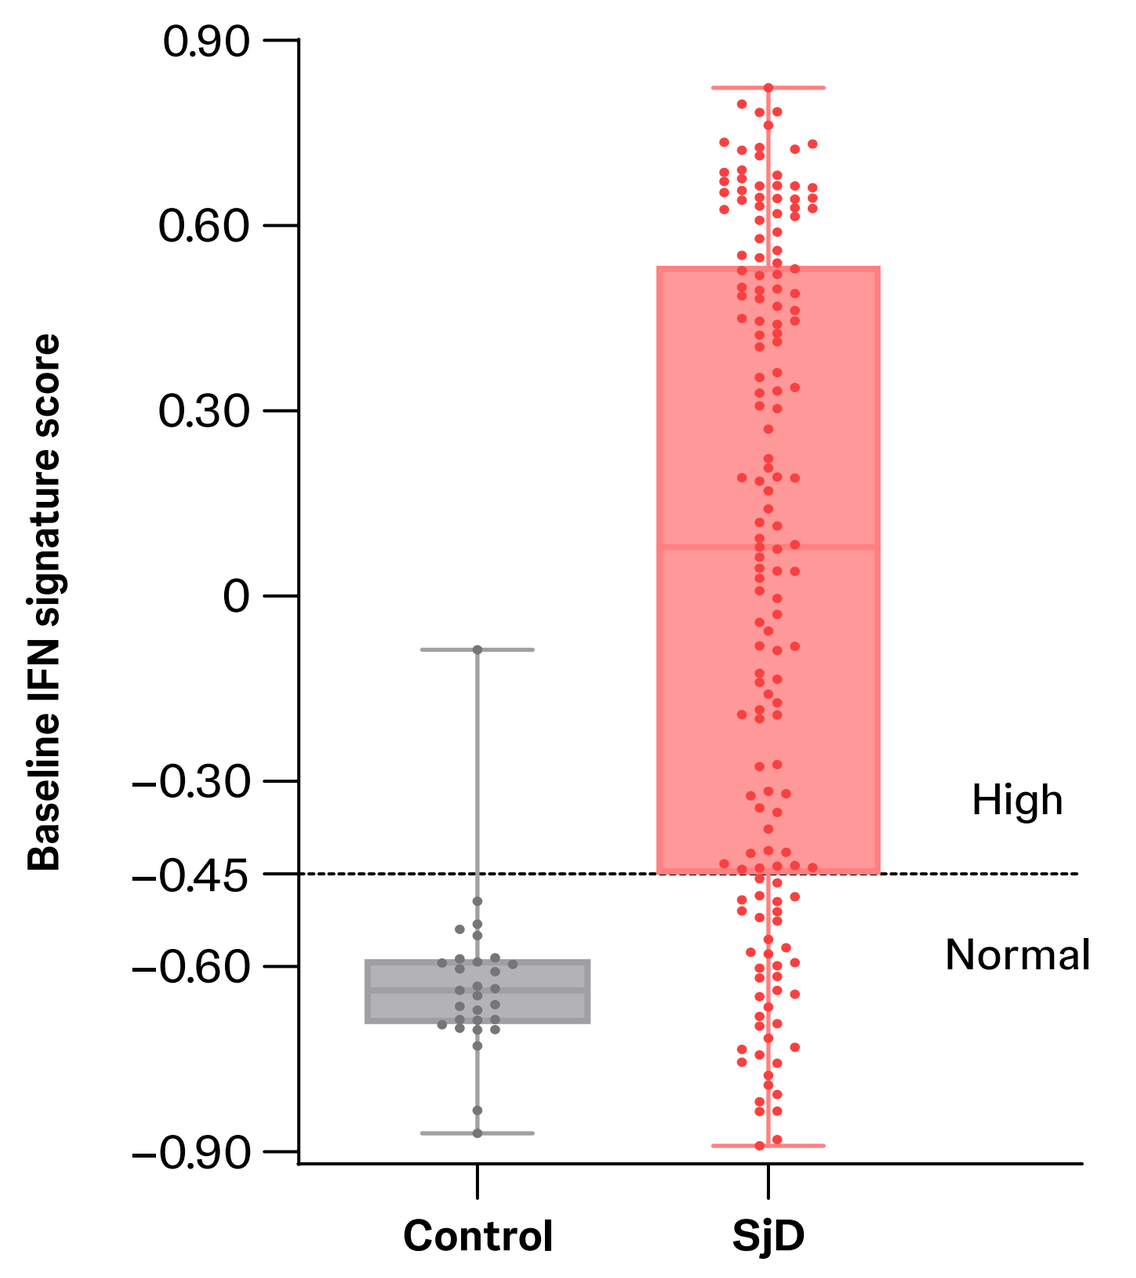
Baseline IFN signature score
High
Normal
Control
SjD

ClinESSDAI-4 responses at Week 24 in IFN-high participants

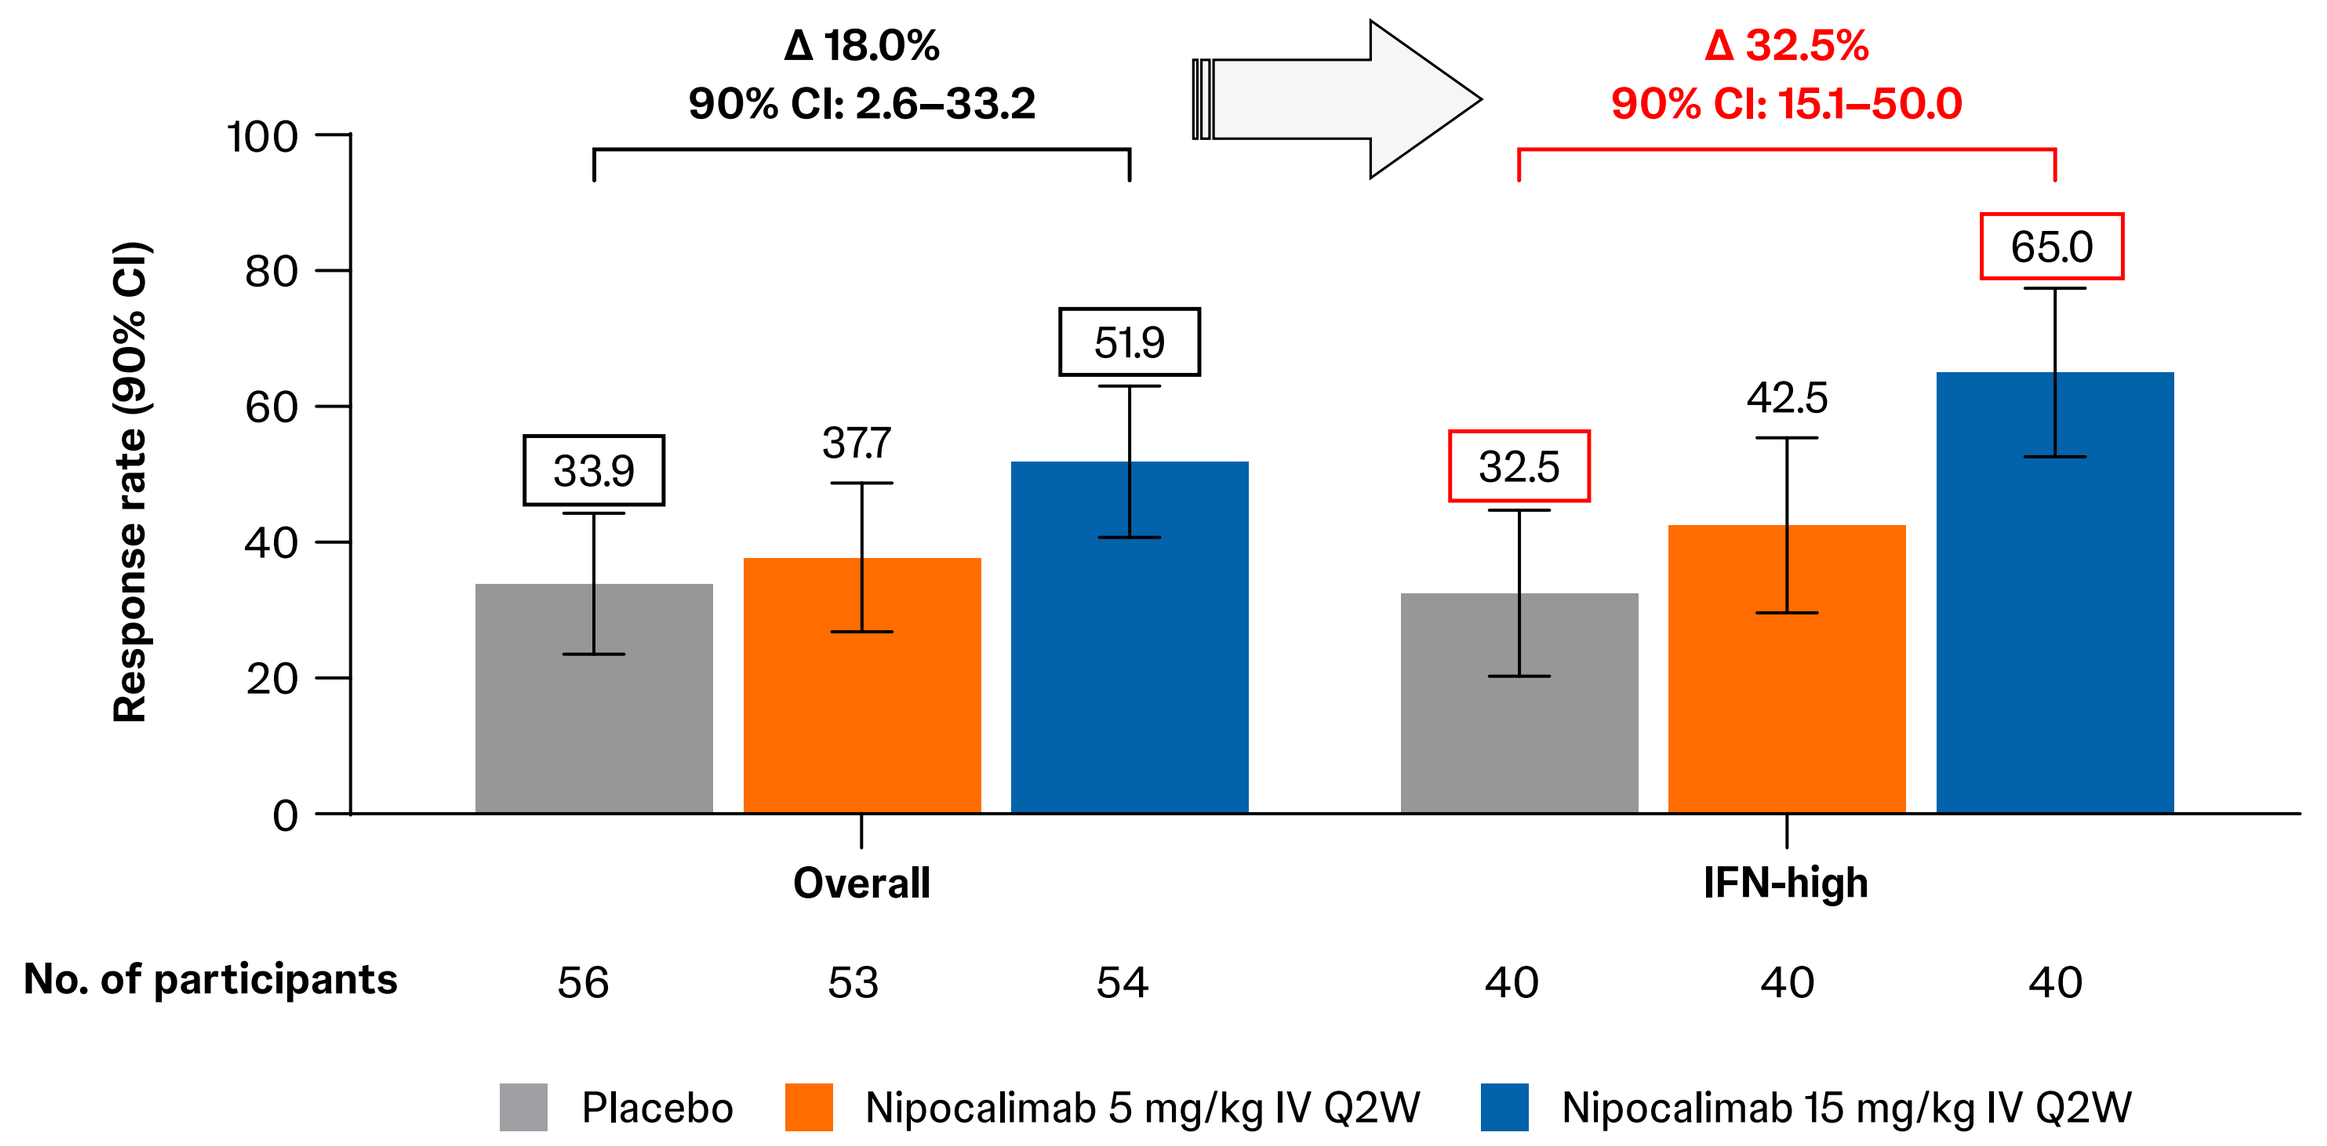
Δ 18.0%
90% CI: 2.6–33.2
Δ 32.5%
90% CI: 15.1–50.0
Response rate (90% CI)
33.9
37.7
51.9
32.5
42.5
65.0
Overall
IFN-high
No. of participants 56 53 54 40 40 40
Placebo
Nipocalimab 5 mg/kg IV Q2W
Nipocalimab 15 mg/kg IV Q2W

CI=confidence interval, ClinESSDAI=Clinical European Alliance of Associations for Rheumatology Sjögren's Syndrome Disease Activity Index, IFN=interferon, IV=intravenous, Q2W=every 2 weeks, SjD=Sjögren's disease.

High Baseline IgG Level Enriches Responders to Nipocalimab

- Enriched nipocalimab response in IgG-high population (75% of DAHLIAS participants)

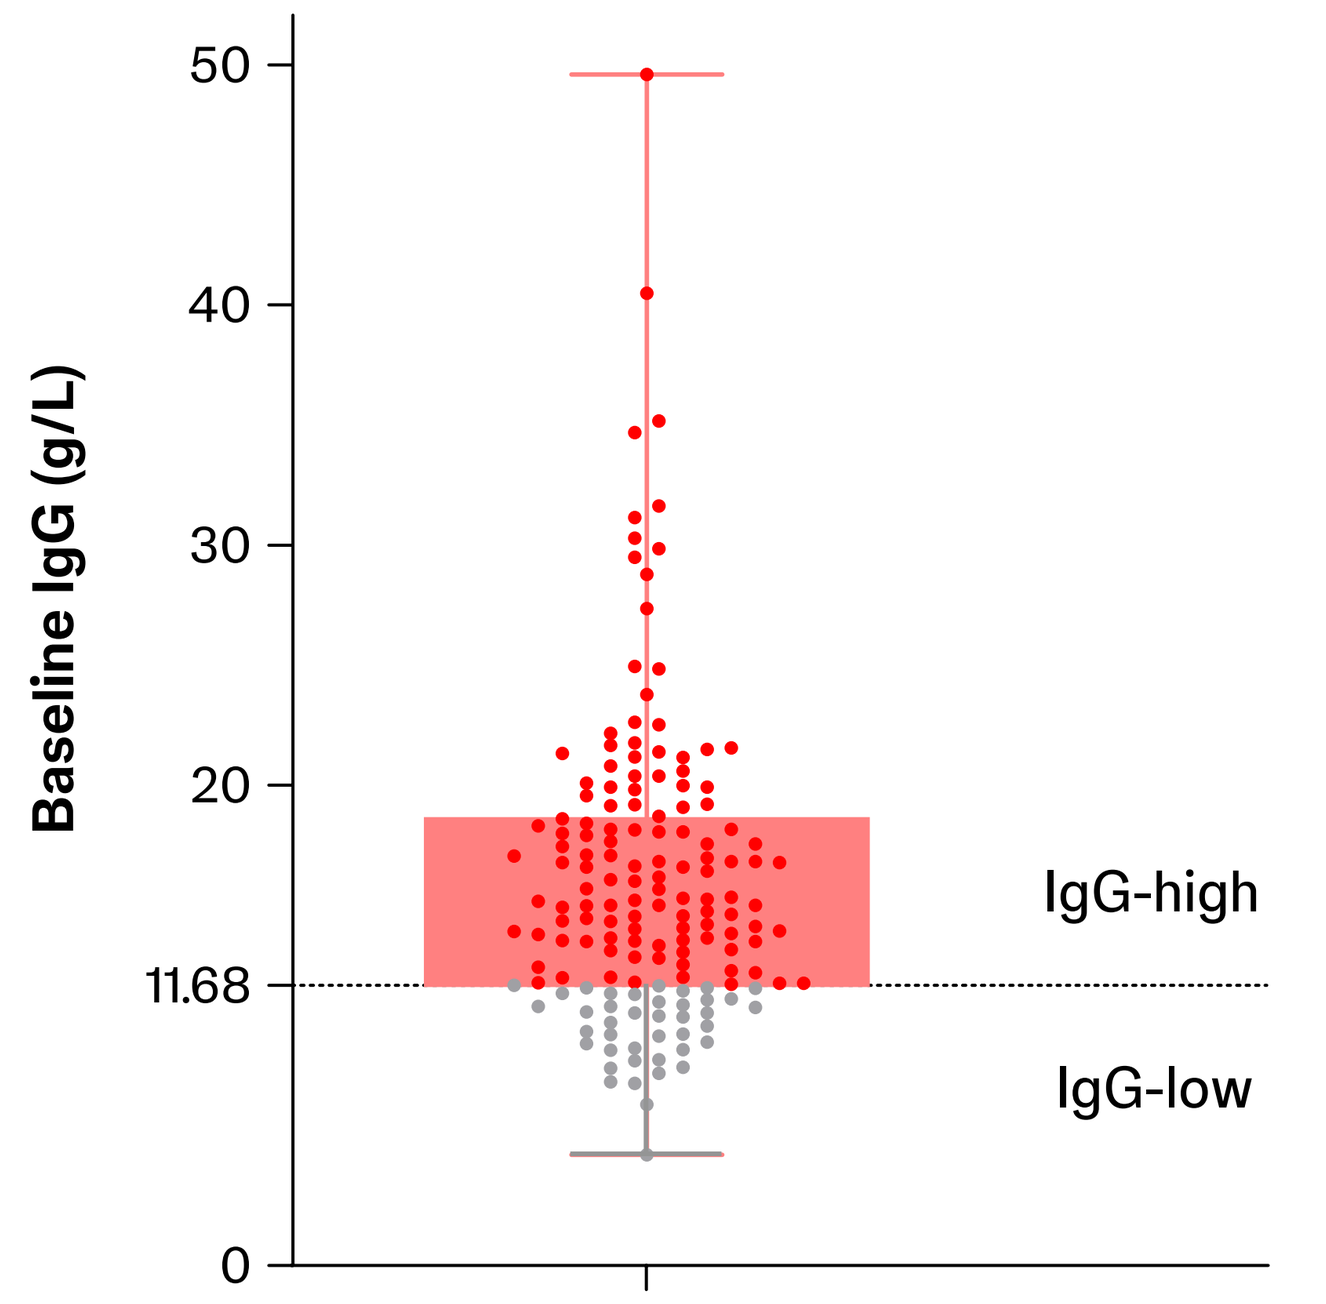
Baseline IgG (g/L)
11.68
IgG-high
IgG-low

ClinESSDAI-4 responses at Week 24 in IgG-high participants

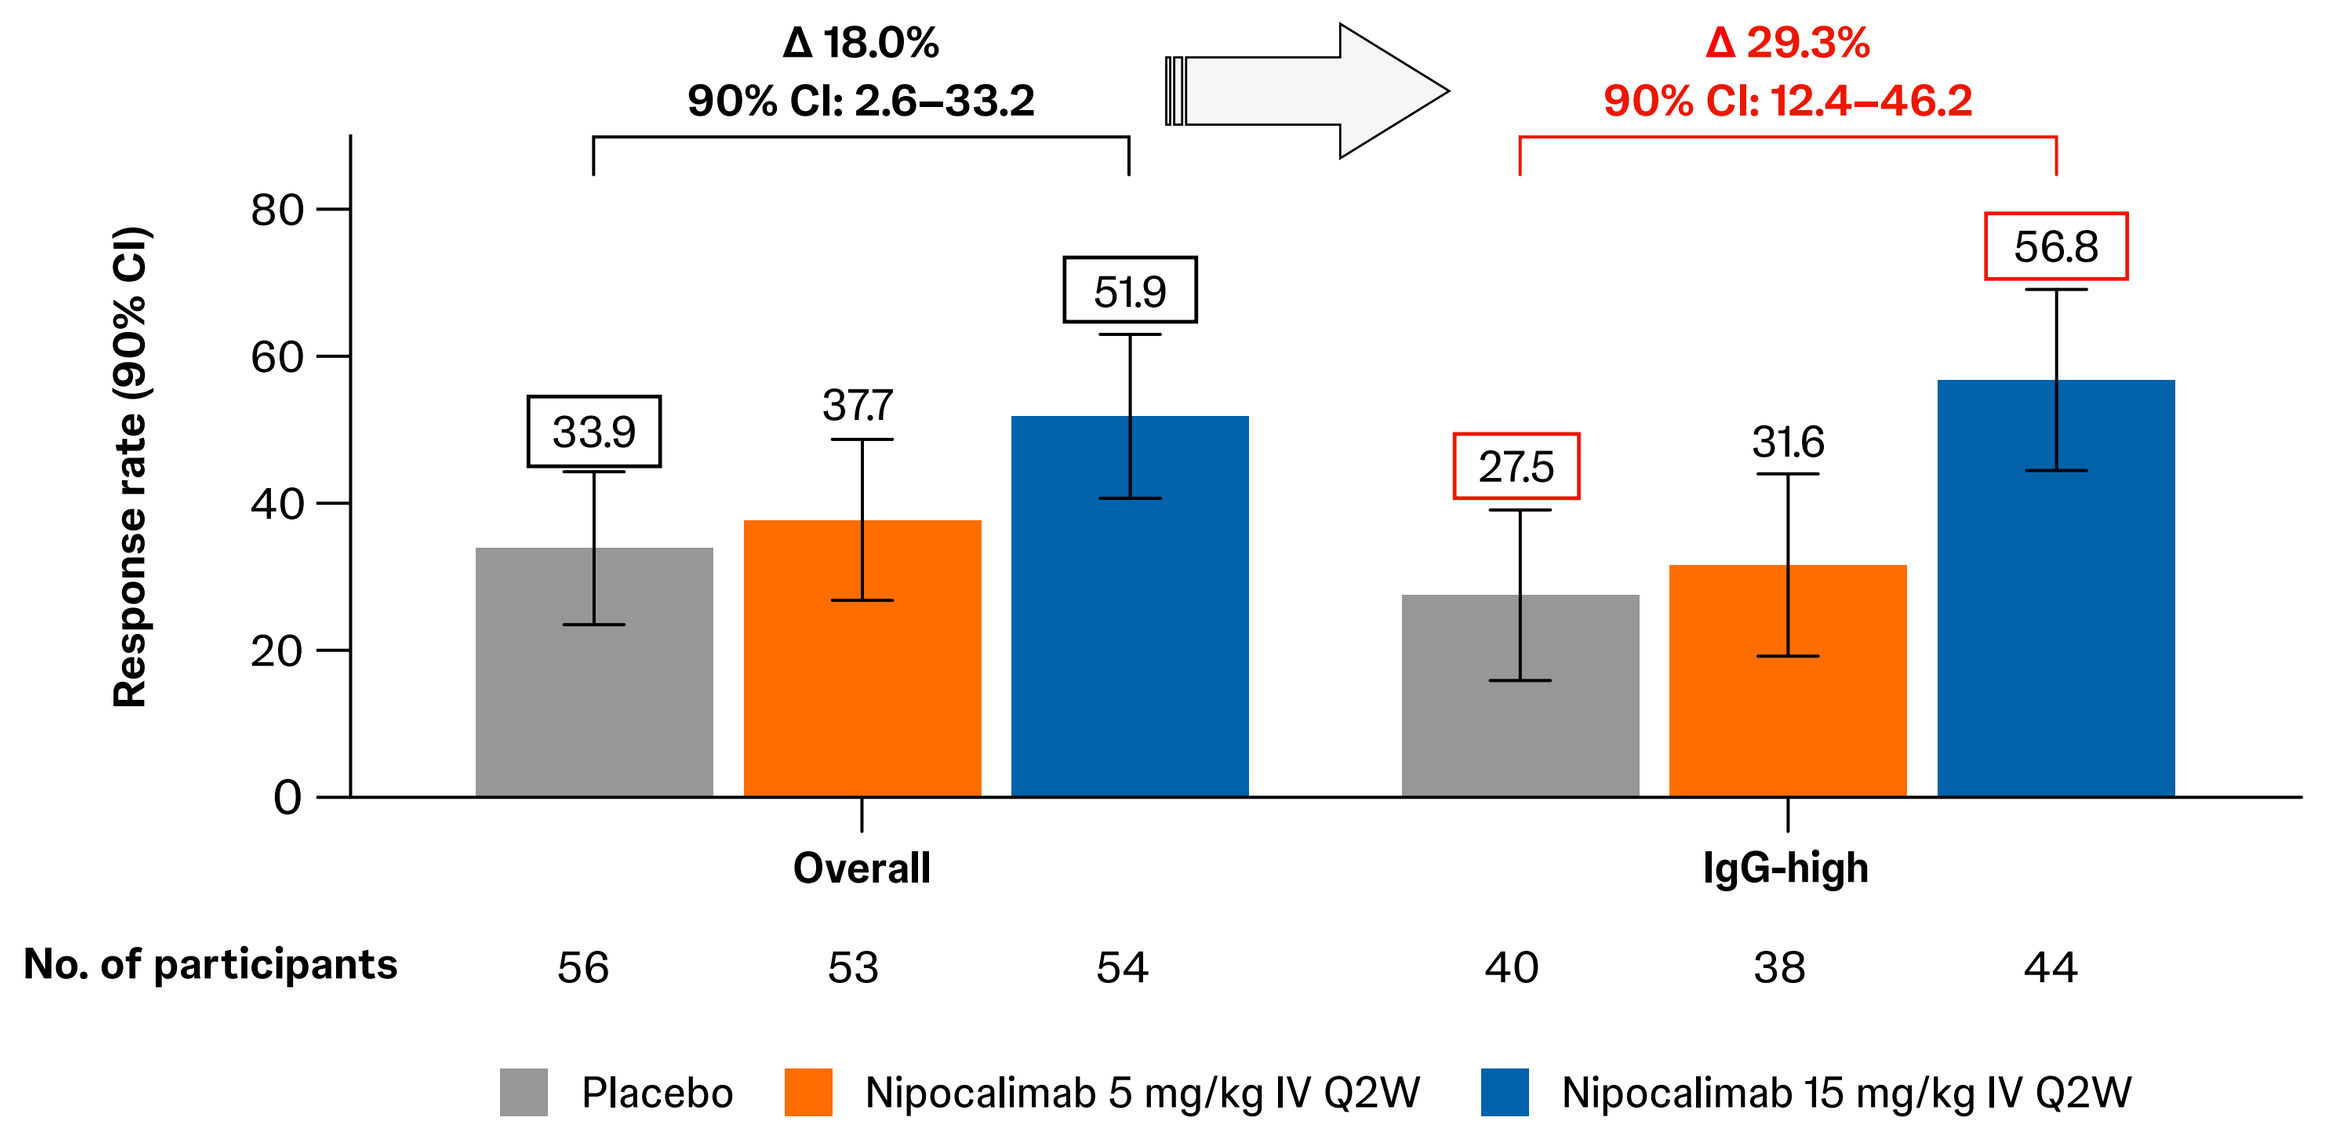
Δ 18.0%
90% CI: 2.6–33.2
Δ 29.3%
90% CI: 12.4–46.2
Response rate (90% CI)
33.9
37.7
51.9
27.5
31.6
56.8
Overall
IgG-high
No. of participants 56 53 54 40 38 44
Placebo
Nipocalimab 5 mg/kg IV Q2W
Nipocalimab 15 mg/kg IV Q2W

CI=confidence interval, ClinESSDAI=Clinical European Alliance of Associations for Rheumatology Sjögren's Syndrome Disease Activity Index, IgG=immunoglobulin G, IV=intravenous, Q2W=every 2 weeks.

PRESENTED BY: Kathleen A. Packard at the Congress of Clinical Rheumatology-West (CCR-West), September 17–20, 2026; Huntington Beach, California, USA.
REFERENCES: 1. Nocturne G, Mariette X. *Nat Rev Rheumatol.* 2013;9(9):544–556. **2.** Mariette X, Criswell LA. *N Engl J Med.* 2018;378(10):931–939. **3.** Beydon M, et al. *Nat Rev Rheumatol.* 2024;20(3):158–169. **4.** Huang H, et al. *Rheumatology (Oxford).* 2021;60(9):4029–4038. **5.** Parisis D, et al. *J Clin Med.* 2020;9(7):2299. **6.** Zhang Y, et al. *J Clin Rheumatol.* 2024;30(4):151–158. **7.** Retamozo S, et al. *Clin Exp Rheumatol.* 2021;39(suppl 133):S166–S174. **8.** Antozzi C, et al. *Neurology.* 2024;102(2):e207937. **9.** Moise KJ Jr, et al. *N Engl J Med.* 2024;391(6):526–537. **10.** Noaiseh G, et al. *Lancet.* 2025;406(10518):2435–2448.
ACKNOWLEDGEMENTS: We thank all DAHLIAS study participants, investigators, and study site staff for their contributions. Medical writing support was provided by Aya Younes, PharmD, of Lumanity Communications Inc., and was funded by Johnson & Johnson. Layout design and formatting for this encore presentation was provided by Ganesh Karpe (SIRO Medical Writing Pvt. Ltd., India). This study and presentation were sponsored by Johnson & Johnson. **DISCLOSURES: HS** received grant funding from the National Institutes of Health (NIH) and the US Department of Veterans Affairs; consulted and participated in advisory boards for IQVIA, Johnson & Johnson, Veloxis Pharmaceuticals, and Vor Bio; and served as a local site investigator for Johnson & Johnson. **J-EG** consulted for AbbVie, Bristol Myers Squibb, Galapagos, Gilead, Johnson & Johnson, Lilly, MSD, Novartis, Pfizer, Sanofi, and UCB. **HL, DG, KM, LT, CS, KAP, ML, JH, KC, DW, SG, CC, SL,** and **KS** are employees of Johnson & Johnson and own stocks or stock options in Johnson & Johnson. **PREVIOUSLY PRESENTED AT:** The European Alliance of Associations for Rheumatology (EULAR) European Congress; June 3–6, 2026; London, England, UK.