Analysis of Healthcare Utilization in Primary and Associated Sjögren's Disease

Nora Sandorfi[1], Alexander Keenan[2], Elizabeth Adamson[2], Federico Zazzetti[2], Soumya D Chakravarty[2,3], Debbie Goldschmidt[4], Sakshi Sethi[4], Jessica Smith[4], Esen K Akpek[5]

[1]University of Pennsylvania, Philadelphia, PA, USA; [2]Johnson & Johnson, Horsham, PA, USA; [3]Drexel University School of Medicine, Philadelphia, PA, USA; [4]Analysis Group, New York, NY, USA; [5]Foster Center for Ocular Immunology and Inflammation, Duke University School of Medicine, Durham, NC, USA

Presenter: Archana Jeyakumar

Scan the QR code.

The QR code is intended to provide scientific information for individual reference, and the information should not be altered or reproduced in any way.

Key Takeaways

- **In this descriptive study, patients with pSjD and those with aSjD both experienced multiple symptoms and comorbidities, resulting in substantial HRU in the year leading up to diagnosis**
- **The pre-diagnosis HRU was broadly similar across both patient groups**
- **Following diagnosis, both cohorts experienced a similar rate of increase in HRU, suggesting that SjD itself contributes to disease burden beyond associated autoimmune comorbidities**

Background

Sjögren's disease (SjD) is a chronic systemic autoimmune disorder characterized by glandular dysfunction and symptoms ranging from mucosal dryness to life-threatening organ involvement[1]

– Over 90% of patients with SjD are female[1]

SjD may be primary (pSjD) or associated (aSjD); aSjD is concomitant with another autoimmune disease, commonly systemic lupus erythematosus (SLE) or rheumatoid arthritis (RA)[1,2]

– Approximately one third of patients with SjD are diagnosed with aSjD[3]

Drivers of healthcare resource utilization (HRU) in SjD remain poorly understood, including the extent to which utilization reflects pSjD versus aSjD

Objective

To assess HRU in female patients with pSjD or aSjD

Methods

Study design: retrospective observational cohort study

Study methods and analysis

- The index date was defined as the date of SjD diagnosis
 - The baseline period was the 12 months before diagnosis
 - The follow-up period was the 12 months after diagnosis
- Patient characteristics and diagnoses were assessed in the baseline period
- HRU was assessed in the baseline and follow-up periods
- Patients were classified as aSjD if they had any of 10 autoimmune conditions[a] commonly associated with SjD, including SLE and RA, before index; otherwise, they were classified as pSjD

US Veterans Health Administration data (January 1, 2015, to August 15, 2025)

- **Female adults (aged ≥18 years) with an initial clinical SjD diagnosis on or after October 1, 2021, and no associated autoimmune conditions (pSjD)**
- **Female adults (aged ≥18 years) with an initial clinical SjD diagnosis on or after October 1, 2021, and any of 10 commonly associated autoimmune conditions (aSjD)[a]**

Additional eligibility criteria:

- A secondary (confirmatory) clinical diagnosis ≥30 days after the first clinical diagnosis
- ≥12 months' continuous enrollment before and after diagnosis
- Available dental data

HRU outcomes assessed:

- Inpatient admissions
- Emergency room admissions
- Outpatient visits
- Specialist visits and types
- Rheumatology or other joint or pain care visits
- Ophthalmology/optometry; other vision care; ear, nose, and throat (ENT); and dental visits
- Number of prescription fills

[a]*The 10 commonly associated autoimmune conditions were RA, SLE, autoimmune thyroid disease (Hashimoto's thyroiditis or Grave's disease), scleroderma/systemic sclerosis, psoriatic arthritis, inflammatory bowel disease, autoimmune hepatitis, myasthenia gravis, systemic vasculitis, and cryoglobulinemia.*

Results

- Overall, 31.9% of patients with aSjD had concomitant RA and 31.9% had concomitant SLE

A total of 3,135 patients with SjD were identified from the data source; 1,326 of these patients were female, 451 of whom had available dental data

Pre-Diagnosis Characteristics	Patients with pSjD (n=285)	Patients with aSjD (n=166)
Demographics		
Age at diagnosis, years, mean (SD)	54.6 (11.6)	54.5 (10.6)
Race/ethnicity, n (%)		
White	121 (42.5)	76 (45.8)
Black or African American	92 (32.3)	61 (36.7)
Hispanic or Latino	32 (11.2)	15 (9.0)
Asian or Pacific Islander	10 (3.5)	1 (0.6)
Other or unknown	30 (10.5)	13 (7.8)
Disease Characteristics		
Charlson Comorbidity Index, mean (SD)	0.7 (1.2)	1.3 (1.2)
Number of relevant diagnoses/symptoms, mean (SD)	3.4 (2.2)	4.2 (2.3)
Most common diagnoses/symptoms before diagnosis,[a] n (%)		
Depression	149 (52.3)	80 (48.2)
Joint pain	140 (49.1)	79 (47.6)
Dry eyes	126 (44.2)	95 (57.2)
Hypertension	105 (36.8)	57 (34.3)
Chronic pain	82 (28.8)	41 (24.7)
Fibromyalgia	68 (23.9)	47 (28.3)
Insomnia	66 (23.2)	25 (15.1)
Ocular findings[b]	48 (16.8)	32 (19.3)
Headache	48 (16.8)	20 (12.0)
Fatigue	27 (9.5)	19 (11.4)
Dry mouth	34 (11.9)	21 (12.7)
RA	0	53 (31.9)
SLE	0	53 (31.9)

[a]*Most common diagnoses and symptoms defined as those occurring in >10% of patients within a cohort. Diagnoses/symptoms were not mutually exclusive; patients could have more than one diagnosis/symptom.* [b]*Ocular findings included sterile corneal ulcer/infiltration, corneal haze/scarring, corneal melt/perforation, uveitis, optic neuropathy/neuritis, episcleritis/scleritis, and orbital inflammation.* ***SD****=standard deviation.*

Both cohorts showed elevated HRU before diagnosis; after diagnosis, both cohorts experienced increases across most HRU categories, particularly in inpatient admissions

Pre-Diagnosis Period

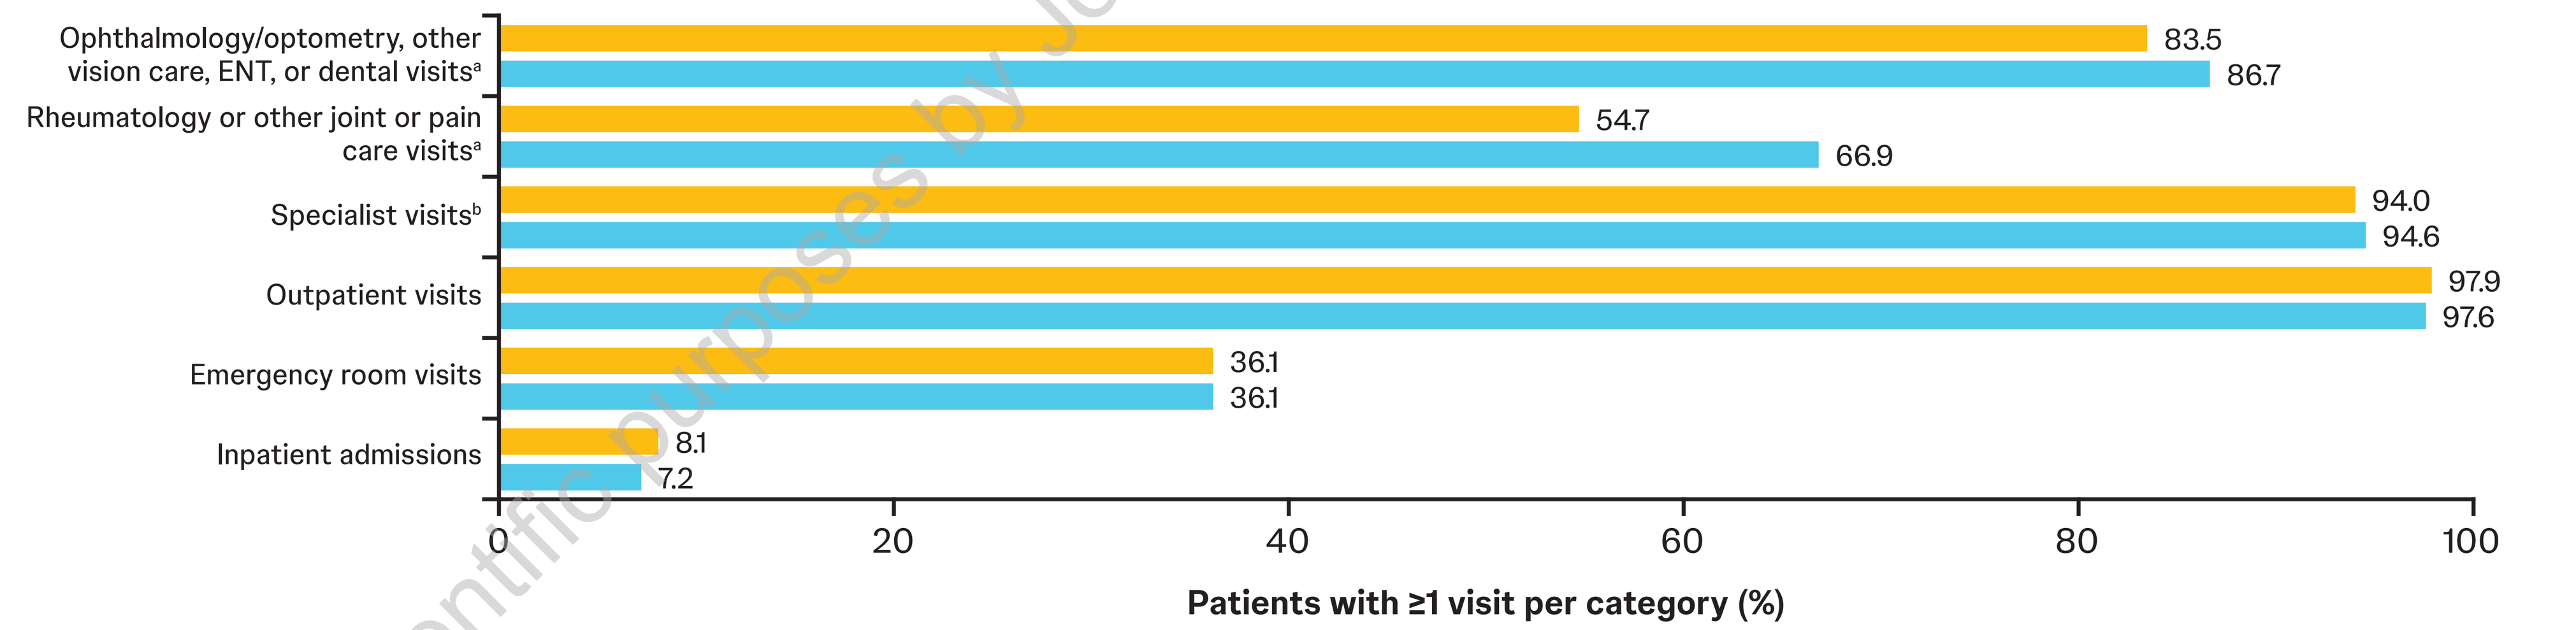
pSjD (n=285)
aSjD (n=166)
Ophthalmology/optometry, other vision care, ENT, or dental visits[a]
83.5
86.7
Rheumatology or other joint or pain care visits[a]
54.7
66.9
Specialist visits[b]
94.0
94.6
Outpatient visits
97.9
97.6
Emergency room visits
36.1
36.1
Inpatient admissions
8.1
7.2
0
20
40
60
80
100
Patients with ≥1 visit per category (%)

Post-Diagnosis Period

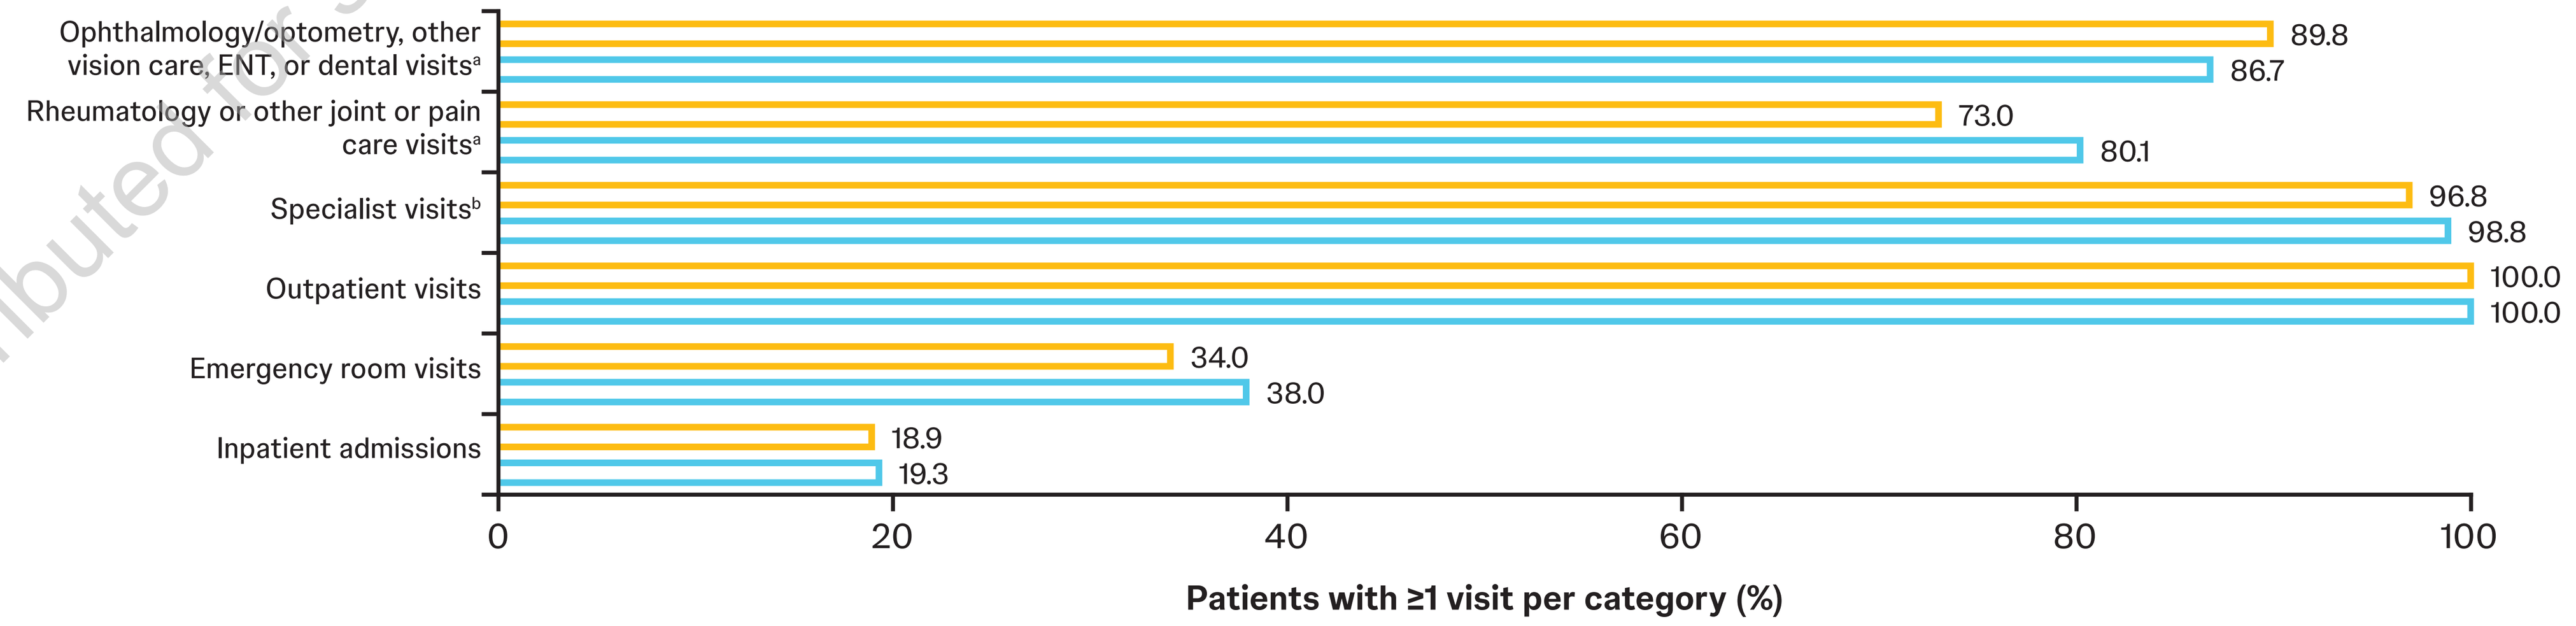
pSjD (n=285)
aSjD (n=166)
Ophthalmology/optometry, other vision care, ENT, or dental visits[a]
89.8
86.7
Rheumatology or other joint or pain care visits[a]
73.0
80.1
Specialist visits[b]
96.8
98.8
Outpatient visits
100.0
100.0
Emergency room visits
34.0
38.0
Inpatient admissions
18.9
19.3
0
20
40
60
80
100
Patients with ≥1 visit per category (%)

[a]*Patients with visits to more than one provider type were counted once in the combined categories.* [b]*Specialist visits included all types of outpatient care, excluding primary care and emergency room visits.*

- After diagnosis, patients with pSjD saw an average of 6.2 specialist types per patient per year (PPPY) and patients with aSjD saw an average of 6.8 specialist types PPPY, representing an incremental increase of 19% for both

The number of specialist types visited PPPY increased after diagnosis

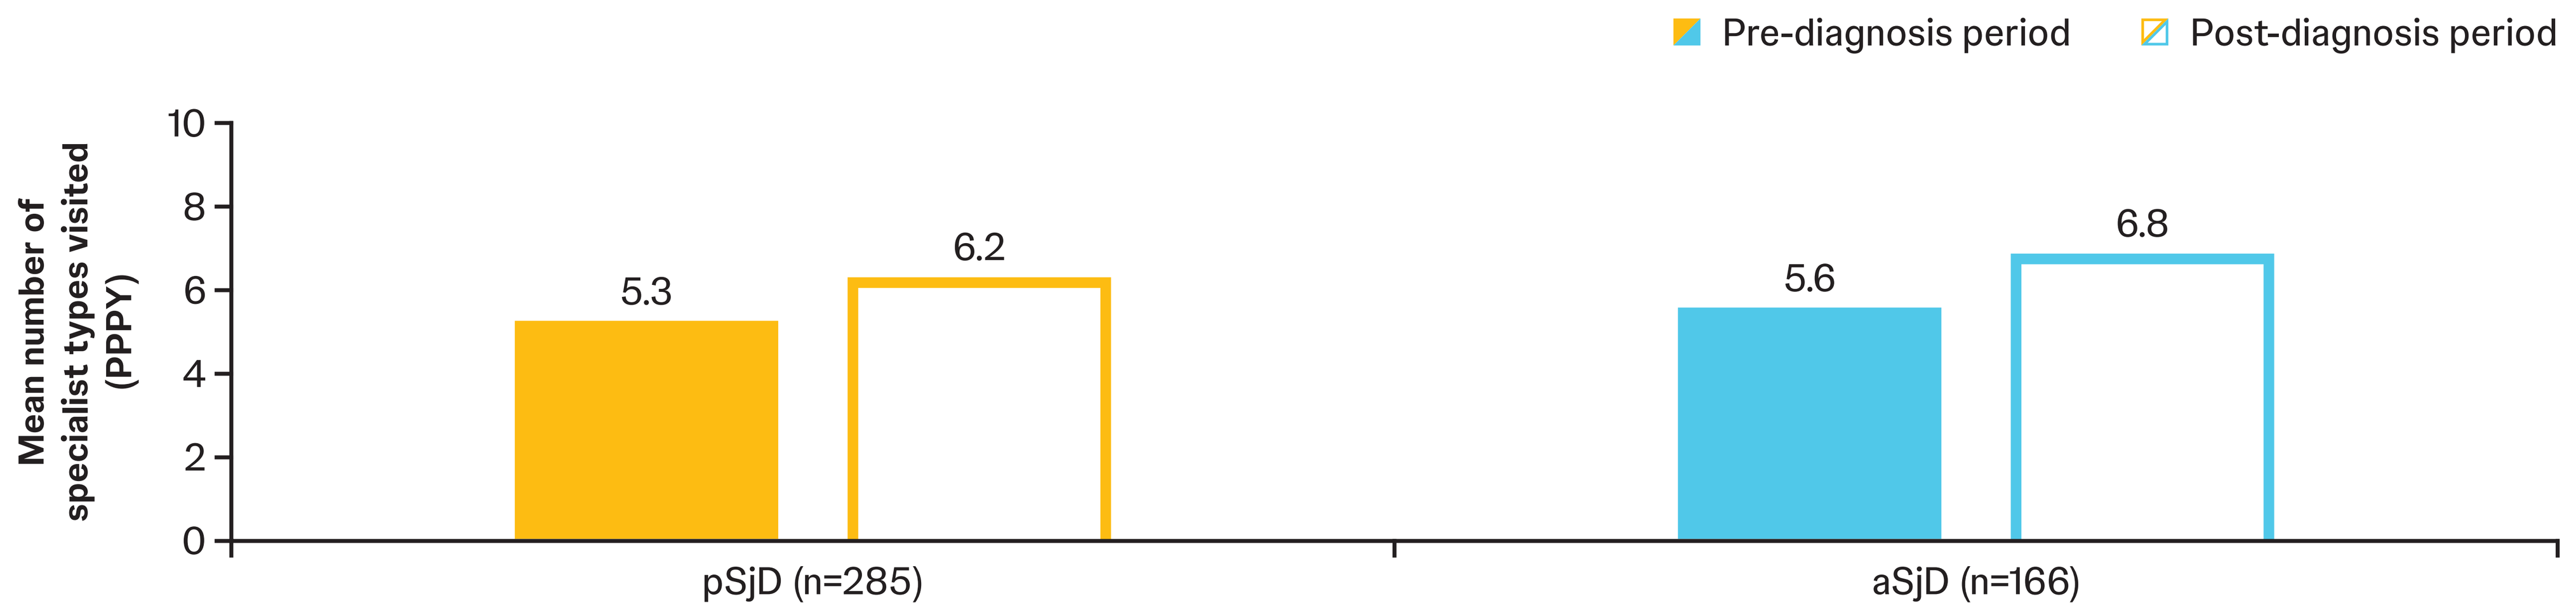
Pre-diagnosis period
Post-diagnosis period
Mean number of specialist types visited (PPPY)
10
8
6
4
2
0
5.3
6.2
5.6
6.8
pSjD (n=285)
aSjD (n=166)

The number of prescription fills PPPY also increased after diagnosis

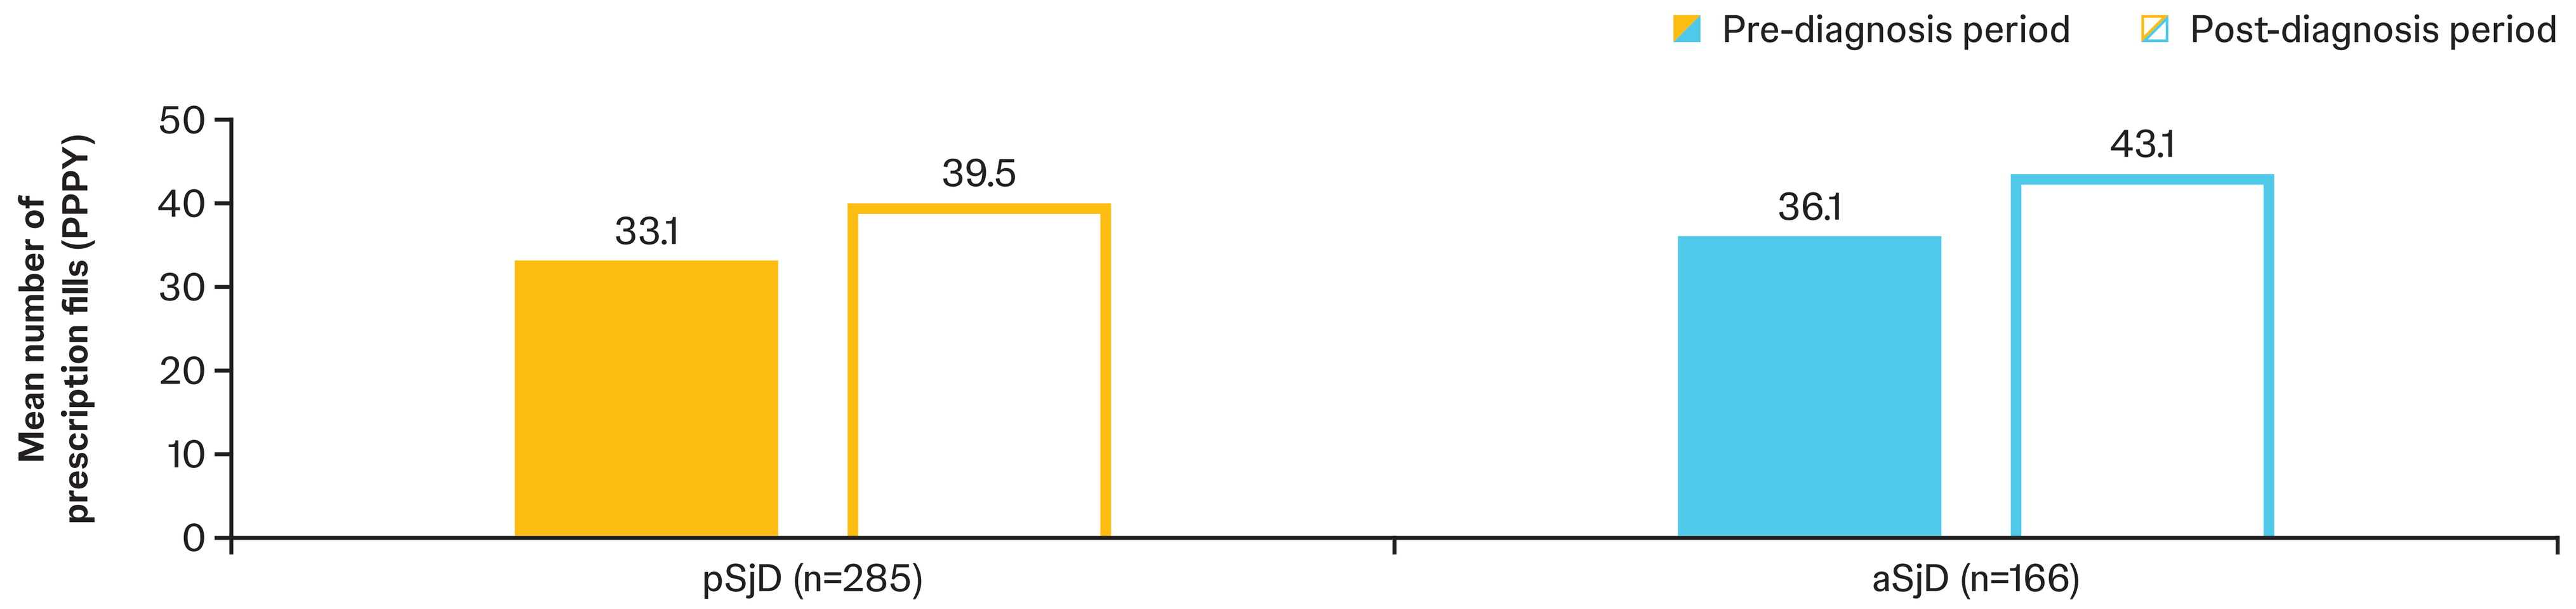
Pre-diagnosis period
Post-diagnosis period
Mean number of prescription fills (PPPY)
50
40
30
20
10
0
33.1
39.5
36.1
43.1
pSjD (n=285)
aSjD (n=166)

PRESENTED AT: The Congress of Clinical Rheumatology (CCR) – West 2026; Huntington Beach, CA, USA; September 17–20, 2026. **REFERENCES: 1.** Beydon M, et al. *Nat Rev Rheumatol.* 2024;20:158–69. **2.** Ramos-Casals M, et al. *Nat Rev Rheumatol.* 2025;21:426–37. **3.** Hernandez-Molina G, et al. *J Rheumatol.* 2010;37:800–8. **ACKNOWLEDGMENTS:** This study was sponsored by Johnson & Johnson. Medical writing support was provided by Allison Michaelis and Adeline Rosenberg on behalf of Twist Medical under the direction of the authors in accordance with Good Publication Practice guidelines (*Ann Intern Med.* 2022;175:1298–304). **DISCLOSURES: NS** has received consulting/advisory board fees from Bristol Myers Squibb, Johnson & Johnson, and Novartis. **AK**, **EA**, **FZ**, **SDC**, and **AJ** (presenter) are employees of and may own stock or stock options in Johnson & Johnson. **DG**, **SS**, and **JS** are employees of Analysis Group, Inc., a consulting company that has received research funding from Johnson & Johnson. **EKA** has received grant/research support from Dompé, the National Institutes of Health, Novartis, Sjögren's Foundation, and the US Department of Defense; and received consulting fees/honoraria from Alcon, Biotissue, Iolyx, Johnson & Johnson, Thea, and W.L. Gore & Associates.

This material is distributed for scientific purposes by Johnson & Johnson Innovative Medicine and is not for promotional use.