Pharmacokinetics of Subcutaneous Ustekinumab in Pediatric Participants With Active Juvenile Psoriatic Arthritis

Thomas N. Kakuda,[1] Nicolino Ruperto,[2,3] Yingyi Li,[4] Wangda Zhou,[4] Jocelyn H. Leu,[4] Lindsey Bozzarello,[4] Sarah Ringold,[1] Samantha Howard,[5] Valerie Smith,[4] Katherine Berezny,[4] Daniel J. Lovell,[6] Alberto Martini,[7] Daniel P. Hawley,[8] Ozgur Kasapcopur,[9] Sandrine Compeyrot-Lacassagne,[10] Hafize Emine Sönmez,[11] Berta Paula Magallares López,[12] Ozge Basaran,[13] Alejandro Brigante,[14] Samundeeswari Deepak,[15] Giovanni Filocamo,[16] Kerstin Nott,[17] Cecilia Marine Santarelli,[18] Anna Gruenpeter,[19] Maria Elena Rama,[20] and Hermine Brunner[6] on behalf of the Pediatric Rheumatology Collaborative Study Group (PRCSG) and Paediatric Rheumatology International Trials Organisation (PRINTO)

[1]Johnson & Johnson, San Diego, CA, USA; [2]Università degli Studi Milano-Bicocca, Milano, Italy; [3]IRCCS Fondazione San Gerardo dei Tintori, PRINTO, Monza, Italy; [4]Johnson & Johnson, Spring House, PA, USA; [5]Johnson & Johnson, High Wycombe, UK; [6]University of Cincinnati, Dept of Pediatrics, Cincinnati Children's Hospital Medical Center, Cincinnati, OH, USA; [7]Università degli Studi di Genova, Genova, Italy; [8]Sheffield Children's Hospital, Sheffield, UK; [9]Istanbul University Cerrahpasa – Cerrahpasa Medical School, Istanbul, Türkiye; [10]Great Ormond Street Hospital for Children and Biomedical Research Centre, London, UK; [11]Kocaeli University Research and Application Hospital, İzmit, Türkiye; [12]Hospital Santa Creu i Sant Pau, Barcelona, Spain; [13]Hacettepe University Department of Pediatric Rheumatology, Ankara, Türkiye; [14]Fundación/Sanatorio Güemes, Buenos Aires, Argentina; [15]Nottingham University Hospitals NHS Trust, Queen's Medical Centre, Nottingham, UK; [16]Fondazione IRCCS Cà Granda Ospedale Maggiore Policlinico and University of Milan, Milan, Italy; [17]University Hospital Southampton, NHS Trust, UK; [18]Centro de Investigaciones Médicas Tucumán, Tucumán, Argentina; [19]Centrum Zdrowia Dziecka I Rodziny im Jana Pawla II w Sosnowcu Sp. z o.o., Sosnowiec, Poland; [20]Hospital de Niños de la Santísima Trinidad, Córdoba, Argentina

Scan the QR code.
The QR code is intended to provide scientific information for individual reference, and the information should not be altered or reproduced in any way.

P1006

Key Takeaways

- **Among pediatric pts with active jPsA, UST was associated with improvement in joint and skin symptoms**
- **A previously developed pediatric PsO popPK model was successfully updated with data from pediatric pts with jPsA**
- **UST PK was similar in pts with jPsA by age and in adults with PsA**
- **These data will be used to support an extrapolation approach to broaden the available treatment options for jPsA in Europe, addressing a significant unmet need**

Background and Objectives

Ustekinumab (UST), a monoclonal antibody to the p40 subunit of interleukin-12/23, has demonstrated efficacy and safety in adults and children with moderate-to-severe plaque psoriasis (PsO), active psoriatic arthritis (PsA), active Crohn's disease, or active ulcerative colitis

The open-label, phase 3 PSUMMIT-Jr study (NCT05083182) evaluated subcutaneous UST pharmacokinetics (PK), efficacy, immunogenicity, and safety in pediatric participants (pts) with active juvenile PsA (jPsA)

PSUMMIT-Jr Study Design

Target Population: Children aged ≥5 to <18 years diagnosed with active jPsA[a] for ≥3 months with inadequate response/intolerance to NSAIDs and/or nonbiologic DMARDs

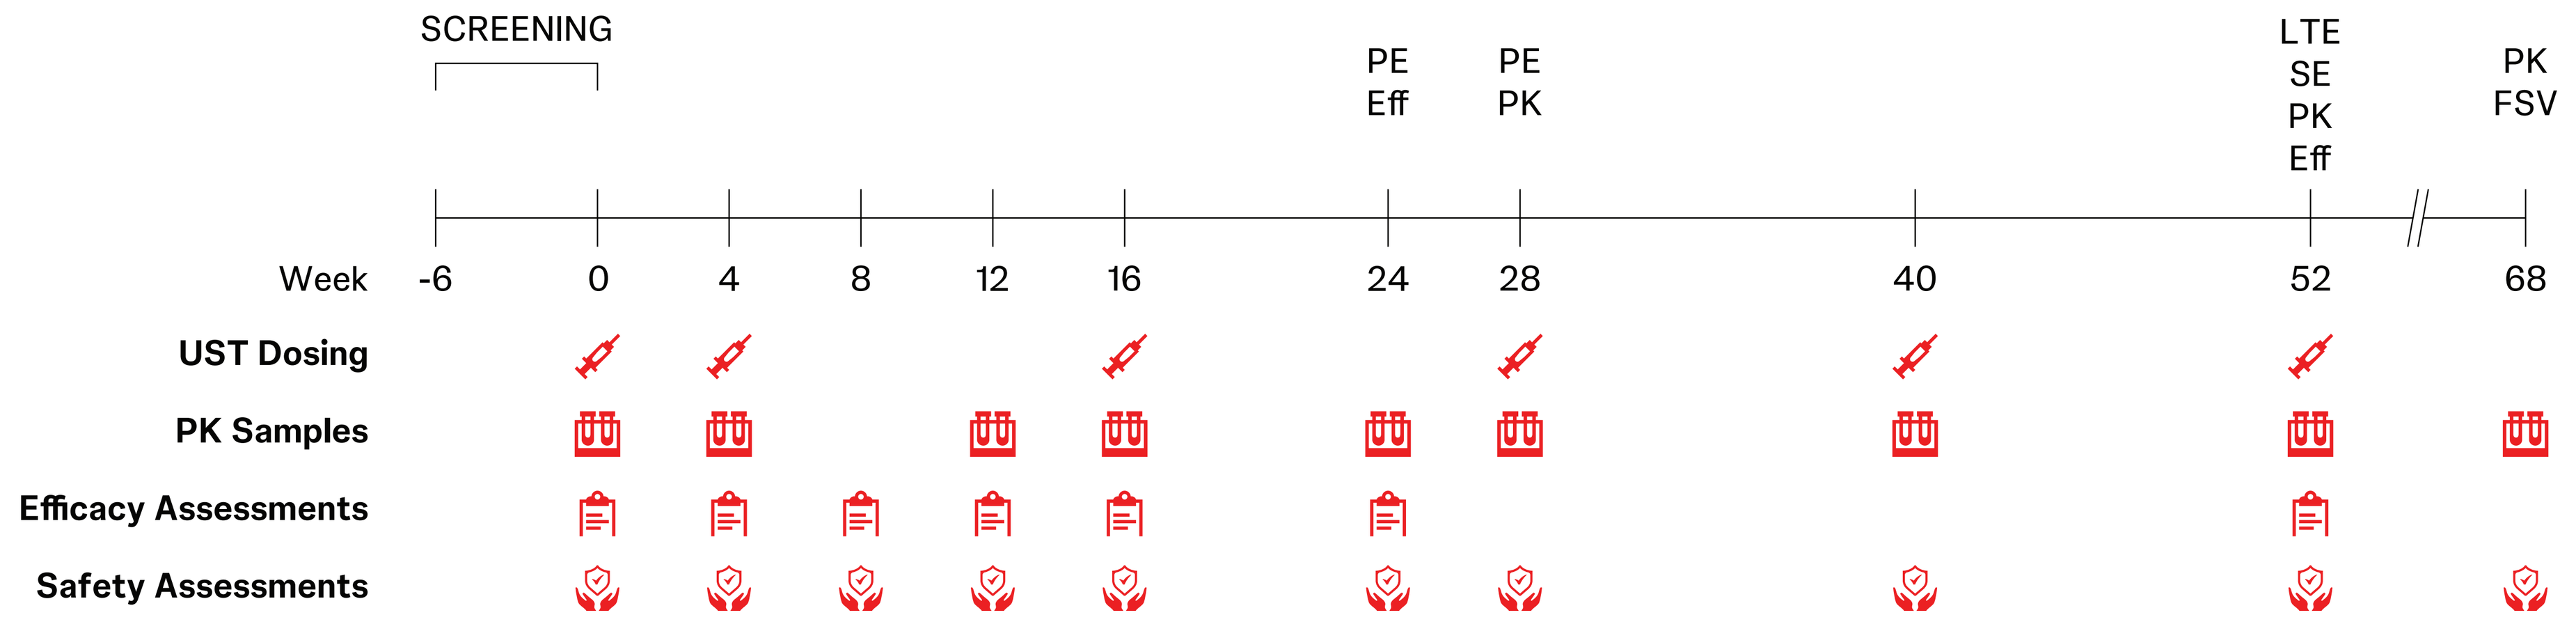
SCREENING
PE Eff
PE PK
LTE SE PK Eff
PK FSV
Week -6 0 4 8 12 16 24 28 40 52 68
UST Dosing
PK Samples
Efficacy Assessments
Safety Assessments

[a]jPsA was defined based on the Vancouver classification criteria: arthritis plus PsO or ≥2 of the following: dactylitis, nail pits, PsO family history, or PsO-like rash. Active disease was defined as ≥3 joints with swelling or loss of motion with pain and/or tenderness. Pts completing the Week-52 visit could enter an LTE study. FSV only occurred for pts not entering the LTE and/or not starting on commercial product; pts starting on commercial product completed the FSV prior to first dose of commercial product. **DMARDs**=disease-modifying anti-rheumatic drugs, **Eff**=efficacy endpoints, **FSV**=final safety visit, **LTE**=long-term extension, **NSAIDs**=nonsteroidal anti-inflammatory drugs, **PE**=primary endpoints, **SE**=secondary endpoints.

Methods

- Pts were assigned to receive subcutaneous UST administered by body weight (<60 kg, 0.75 mg/kg; ≥60 to ≤100 kg, 45 mg; >100 kg, 90 mg)
- Serum UST concentrations were determined using a validated electrochemiluminescence immunoassay (ECLIA) method with a lower quantification limit of 0.1688 μg/mL
- A previously developed population PK (popPK) model (one-compartment linear model with first-order absorption and first-order elimination) for pediatric PsO was used as the starting point for this analysis[1]
- Goodness-of-fit plots and prediction-corrected visual predictive check (VPC) plots were used as external evaluation methods

PK Endpoints

- Steady-state serum trough concentrations ($C_{trough,ss}$) and model-predicted steady-state area under the serum concentration-time curve (AUC_{ss}) over a 12-week dosing interval at Week 28 (primary) and Week 52 (secondary) by age group
- A tertiary endpoint was to compare $C_{trough,ss}$ and AUC_{ss} between children with jPsA (PSUMMIT-Jr) and adults with PsA (PSUMMIT-1 [NCT01009086] and PSUMMIT-2 [NCT01077362])

Results

- All 18 pts enrolled in PSUMMIT-Jr and treated with UST completed the Week-52 study visit

Baseline Demographics and Prior and Current jPsA and PsO Medication Exposure

	UST (N=18)
Demographics	
Age, yrs, median (IQR)	13.0 (11.0; 16.0)
Female	56%
Race	
White	94%
Asian	6%
Weight, kg, median (IQR)	52.1 (41.9; 63.3)
<60	72%
≥60 to 100	28%
Disease Activity	
cJADAS-10, median (IQR)[a]	18.7 (16.4, 21.4)
JADAS-10, median (IQR)[b]	20.5 (13.9, 22.6)
JADAS-27, median (IQR)[b]	16.3 (11.8, 23.3)
JADAS-71, median (IQR)[b]	20.9 (13.9, 26.6)
PGA of PsO (0-4)[c]	2.7 (0.68)
% BSA with PsO[c]	13.2 (12.01)
PASI (0-72)[c]	4.3 (4.29)
jPsA/PsO Medication Use at Baseline	
Nonbiologic DMARDs[d]	61%
Oral corticosteroids	17%
Prednisone or equivalent dose[e]	6.3 (3.2)
NSAIDs	50%
Prior jPsA/PsO Medication Use	
Nonbiologic DMARDs	89%
NSAIDs	78%
Anti-TNFα	56%
Biologics (excluding anti-TNFα)	6%
Janus kinase inhibitors	6%

Data are mean (SD) unless otherwise noted. [a]N=17. [b]N=16. [c]Evaluated among pts with ≥3% BSA involvement and PGA of PsO score ≥2 (mild) at baseline, N=10. [d]Methotrexate. [e]N=3. **BSA**=body surface area, **cJADAS**=clinical Juvenile Arthritis Disease Activity Score, **IQR**=interquartile range, **JADAS**=clinical Juvenile Arthritis Disease Activity Score, **PASI**=Psoriasis Area and Severity Index, **PGA**=Physician Global Assessment, **SD**=standard deviation, **TNF**=tumor necrosis factor.

As previously reported[2]

- Juvenile Idiopathic Arthritis American College of Rheumatology (JIA-ACR) 30/50/70 response rates were 89%/83%/61% at Week 24 and 78%/78%/61% at Week 52 (**Figure**)
- Among pts with ≥3% BSA involvement and PGA of PsO score ≥2 (mild) at baseline, mean PASI score consistently improved (decreased) from baseline (-3.8 and -4.0 at Weeks 24 and 52, respectively)
- No new safety signals were identified
- One pt developed neutralizing antibodies to UST without impact on efficacy endpoints and safety

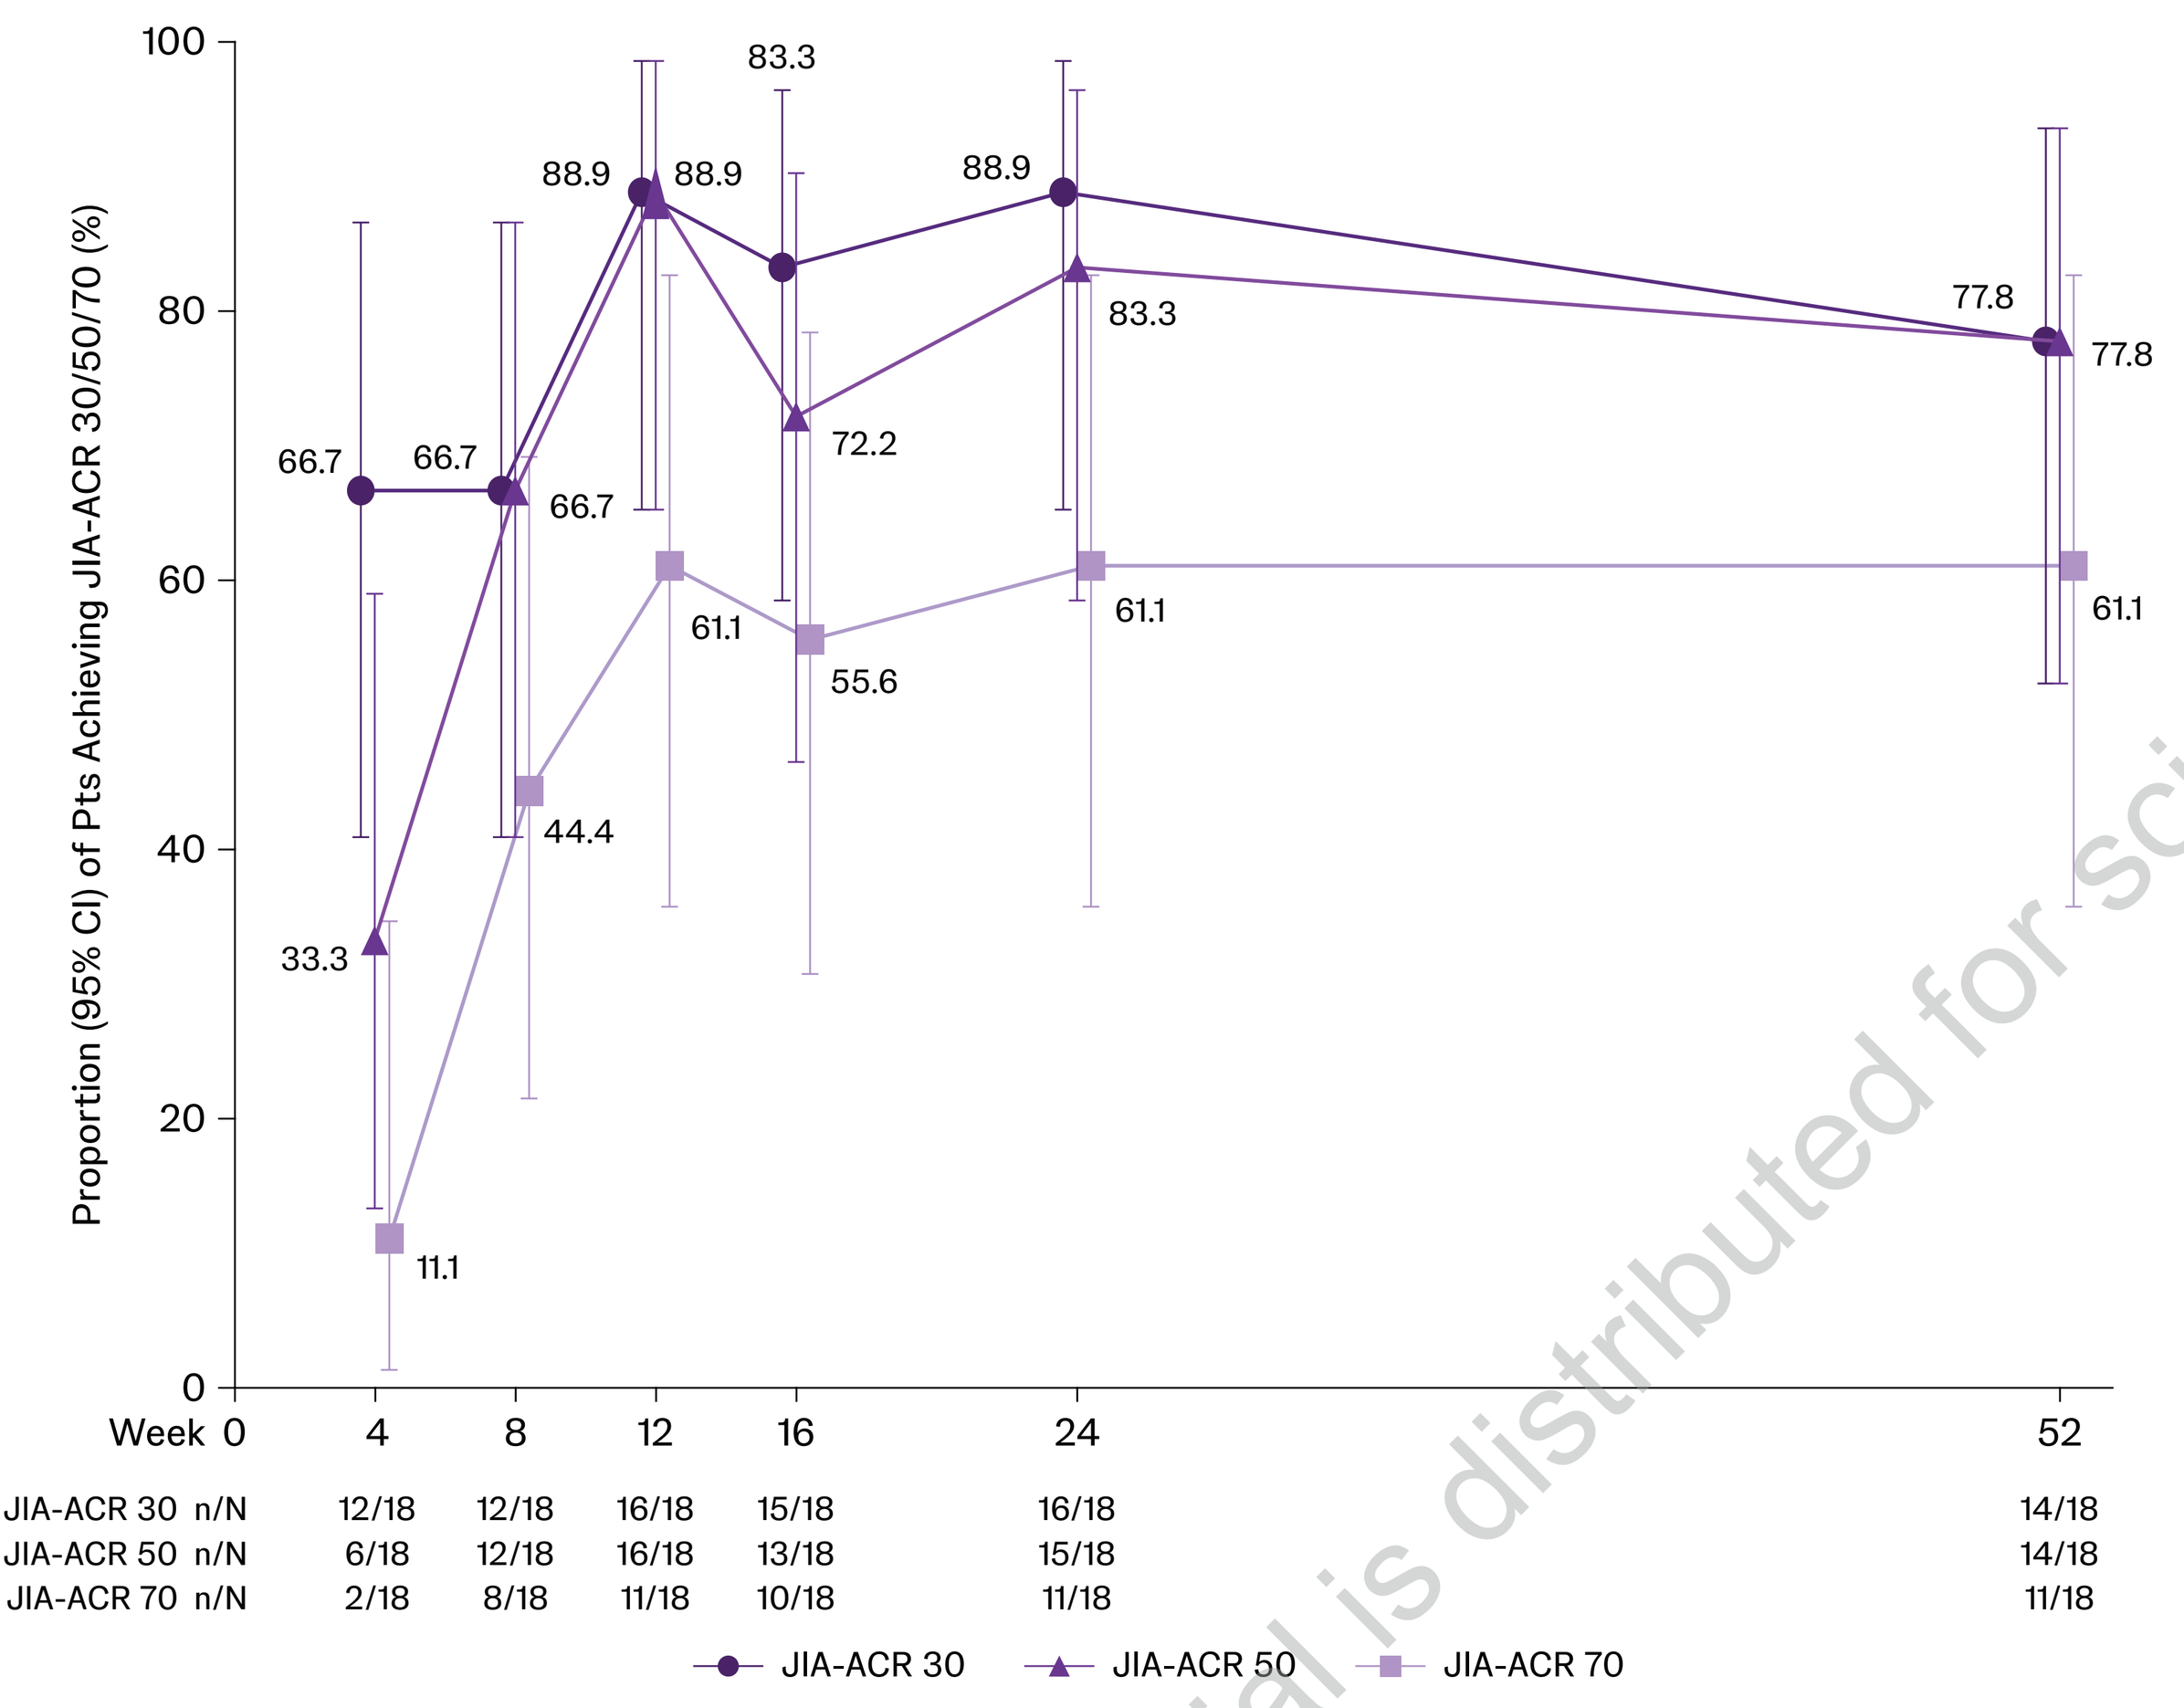
Proportion (95% CI) of Pts Achieving JIA-ACR 30/50/70 (%)
100
80
60
40
20
0
Week 0 4 8 12 16 24 52
66.7
66.7
66.7
33.3
11.1
44.4
88.9
88.9
83.3
72.2
61.1
55.6
88.9
83.3
61.1
77.8
77.8
61.1
JIA-ACR 30 n/N 12/18 12/18 16/18 15/18 16/18 14/18
JIA-ACR 50 n/N 6/18 12/18 16/18 13/18 15/18 14/18
JIA-ACR 70 n/N 2/18 8/18 11/18 10/18 11/18 11/18
JIA-ACR 30
JIA-ACR 50
JIA-ACR 70

JIA-ACR 30/50/70 were defined as 30%/50%/70% improvement from baseline in ≥3 of the following components with worsening of ≥30% in no more than 1 component (PGA of jPsA disease activity, parent/pt assessment of overall well-being, number of active joints, number of joints with limited range of motion, physical function by Childhood Health Assessment Questionnaire, CRP). 95% CIs were calculated based on the Clopper-Pearson method. **CI**=confidence intervals, **CRP**=C-reactive protein.

Goodness-of-fit and prediction-corrected VPC plots confirmed that the central tendency and variability of pediatric UST PK were adequately described in the pediatric PsO popPK model

Goodness-of-fit Plots

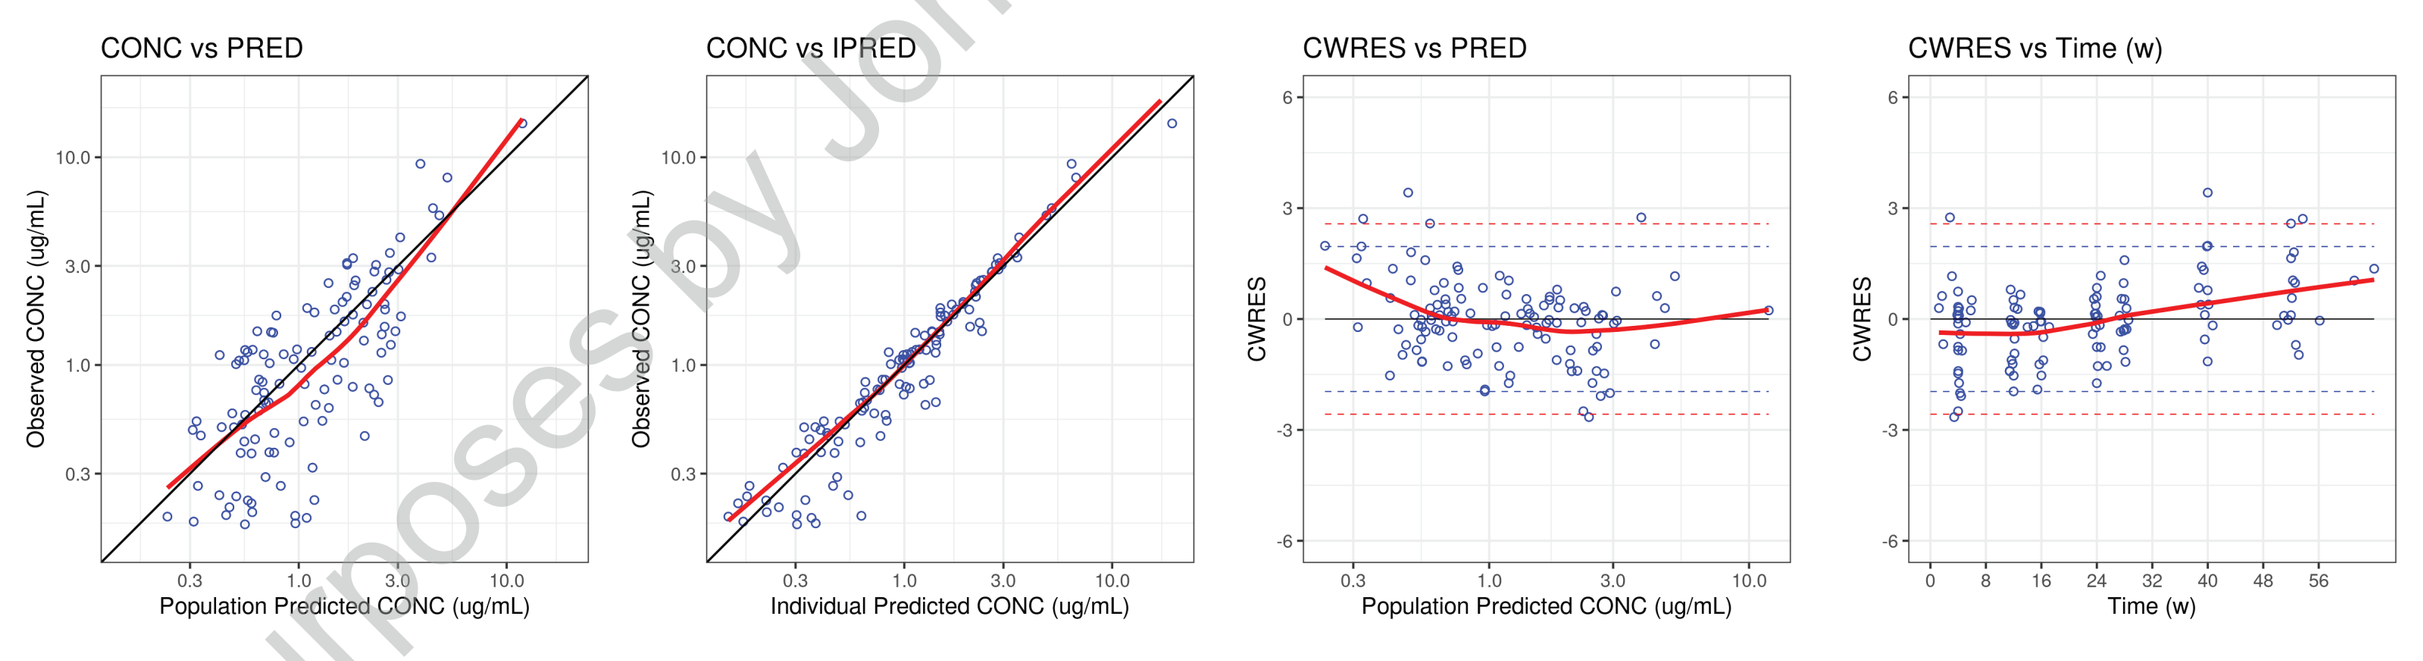
CONC vs PRED
Observed CONC (ug/mL)
Population Predicted CONC (ug/mL)
CONC vs IPRED
Individual Predicted CONC (ug/mL)
CWRES vs PRED
CWRES
CWRES vs Time (w)
Time (w)

Black solid line is the line of identity or the 0 line. Red solid line is the trend line. Blue open circles represent observations. Red dashed lines represent the 99% CI. Blue dashed lines represent the 95% CI. Observations and predictions are presented on logarithmic scale. **CI**=confidence interval; **CONC**=concentration; **CWRES**=conditional weighted residuals; **IPRED**=individual prediction; **PRED**=population prediction; **w**=week.

Prediction-corrected VPC Plots

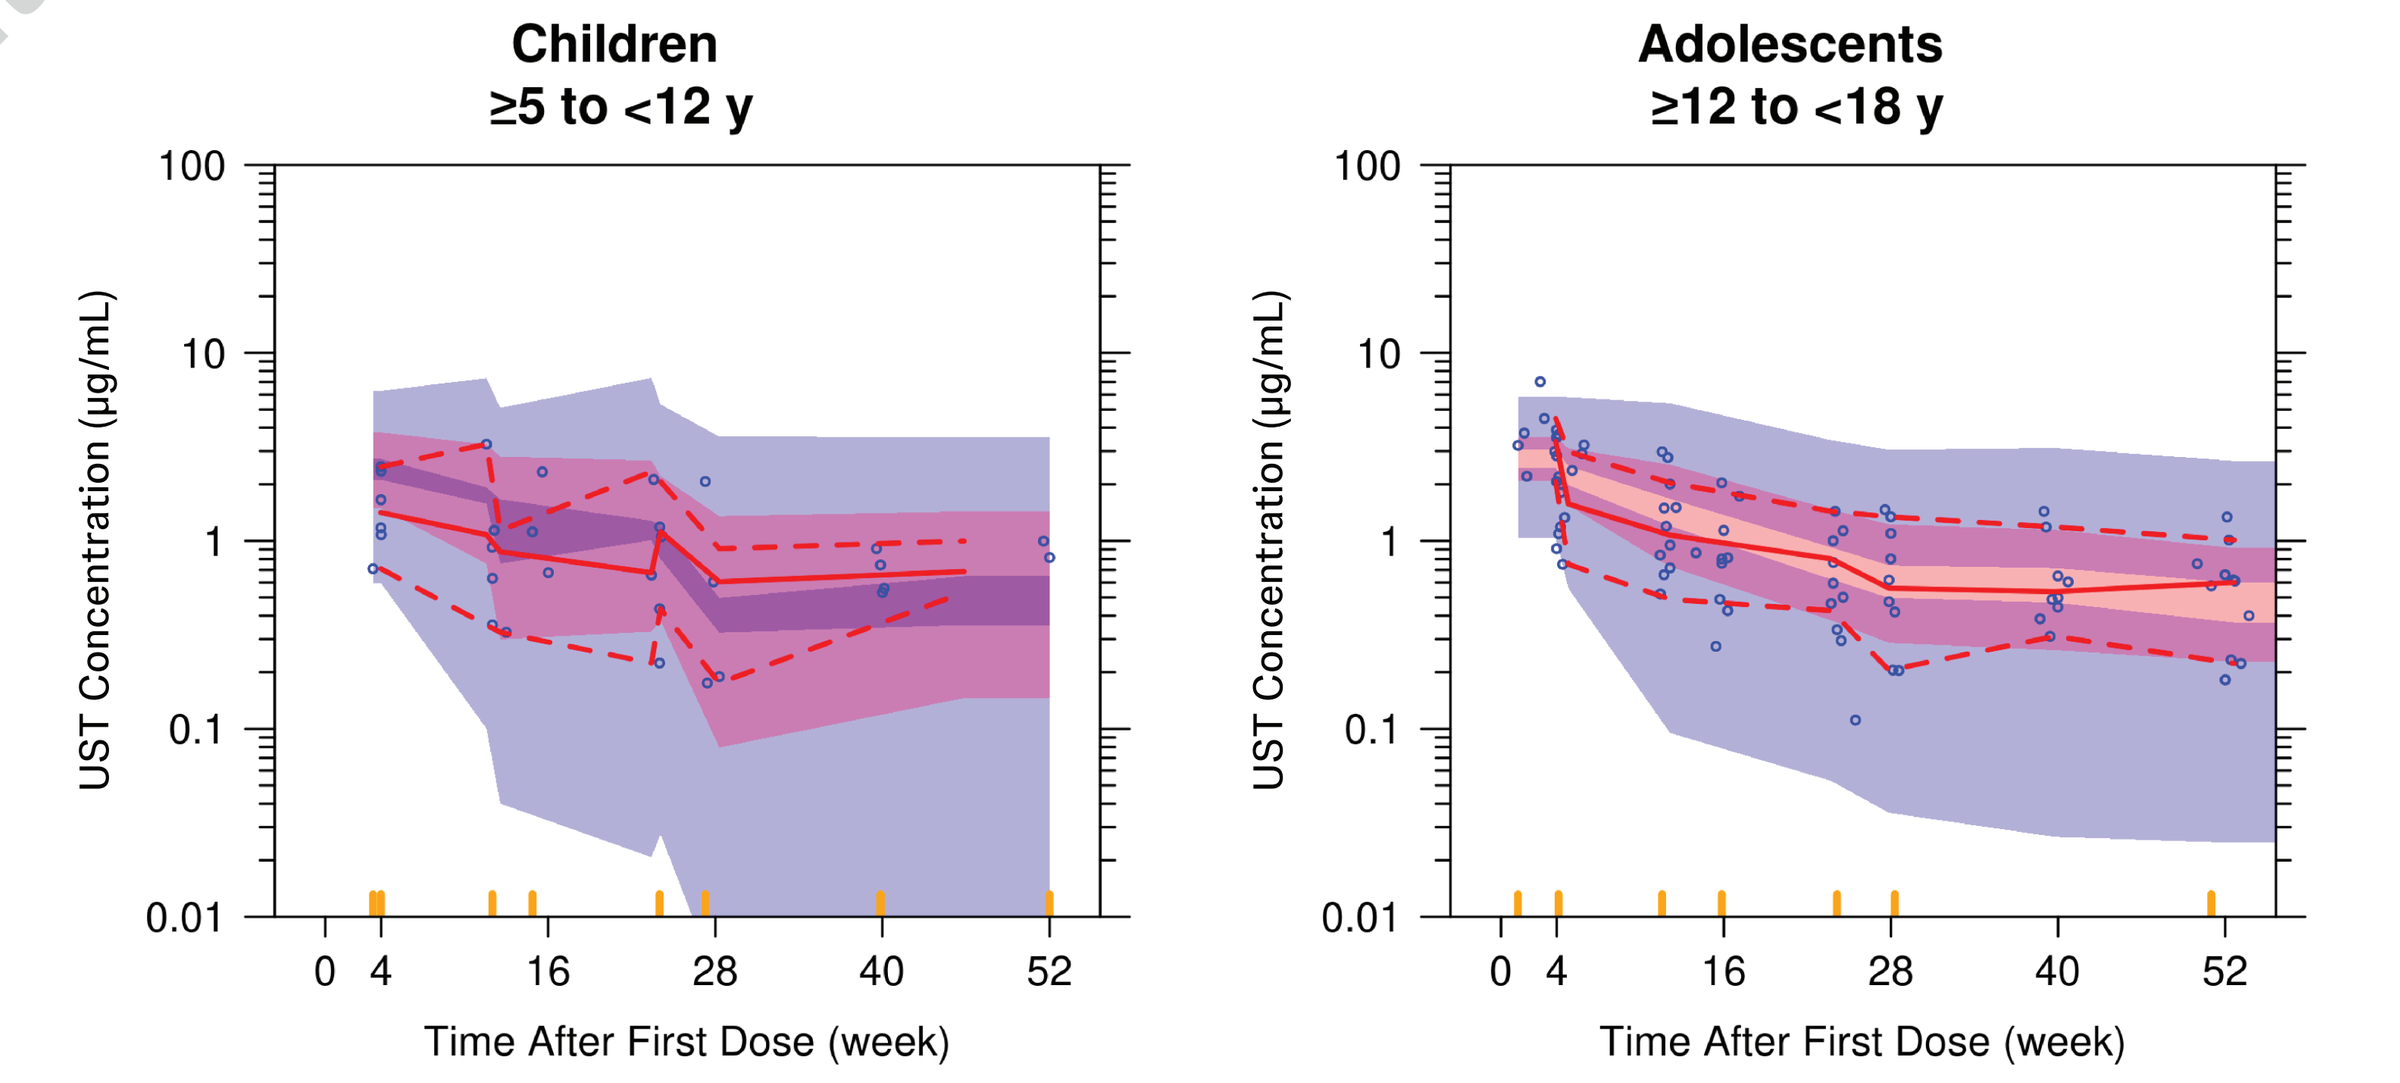
Children ≥5 to <12 y
Adolescents ≥12 to <18 y
UST Concentration (μg/mL)
100
10
1
0.1
0.01
0 4 16 28 40 52
Time After First Dose (week)

The blue open circles represent observed concentrations, the solid lines represent the median, and the dashed lines represent the 5th and 95th percentiles of the observed values. The red shaded areas represent the 95% CI of the median and the blue shaded areas represent the 95% CI of the 5th and 95th percentiles of the simulated concentrations based on 1,000 PopPK simulations. The yellow ticks represent the bins. **y**=years.

Typical UST parameter estimates from the previously developed pediatric PsO popPK model in a child weighing 56 kg

Parameter (Units)	Estimate	RSE (%)	IIV (%CV)	RSE (%)	Shrinkage (%)
CL/F (L/day)	0.204	4.53	38.9	10.9	5.83
V/F (L)	6.77	4.12	30.2	14.2	26.3
k_a (1/day)	0.371	30.2			
IRP on CL	1.32	9.02			
Prop Err (%CV)	24.2	10.0			

Body weight effect exponents on CL/F and V/F were fixed to 0.75 and 1, respectively. The covariance between CL/F and V/F was 40.1%, and the RSE was 41.5%. CL/F (L/day)=0.204*(BWT/56)^0.75*1.32^IRP*exp (ηCL). V/F (L)=6.77*(BWT/56)^1.0*exp (ηV). **%CV**=percentage coefficient of variation; **BWT**=baseline body weight in kilograms; **CL**=clearance; **CL/F**=apparent clearance; **IIV**=inter-individual variability; **IRP**=immune response positive (positive=1, negative=0); k_a=absorption rate constant; **Prop Err**=proportional error; **RSE**=relative standard error; **V/F**=apparent volume of distribution.

Observed $C_{trough,ss}$ and model-predicted AUC_{ss} were comparable among children and adolescents with jPsA and adults with PsA at Weeks 28 and 52

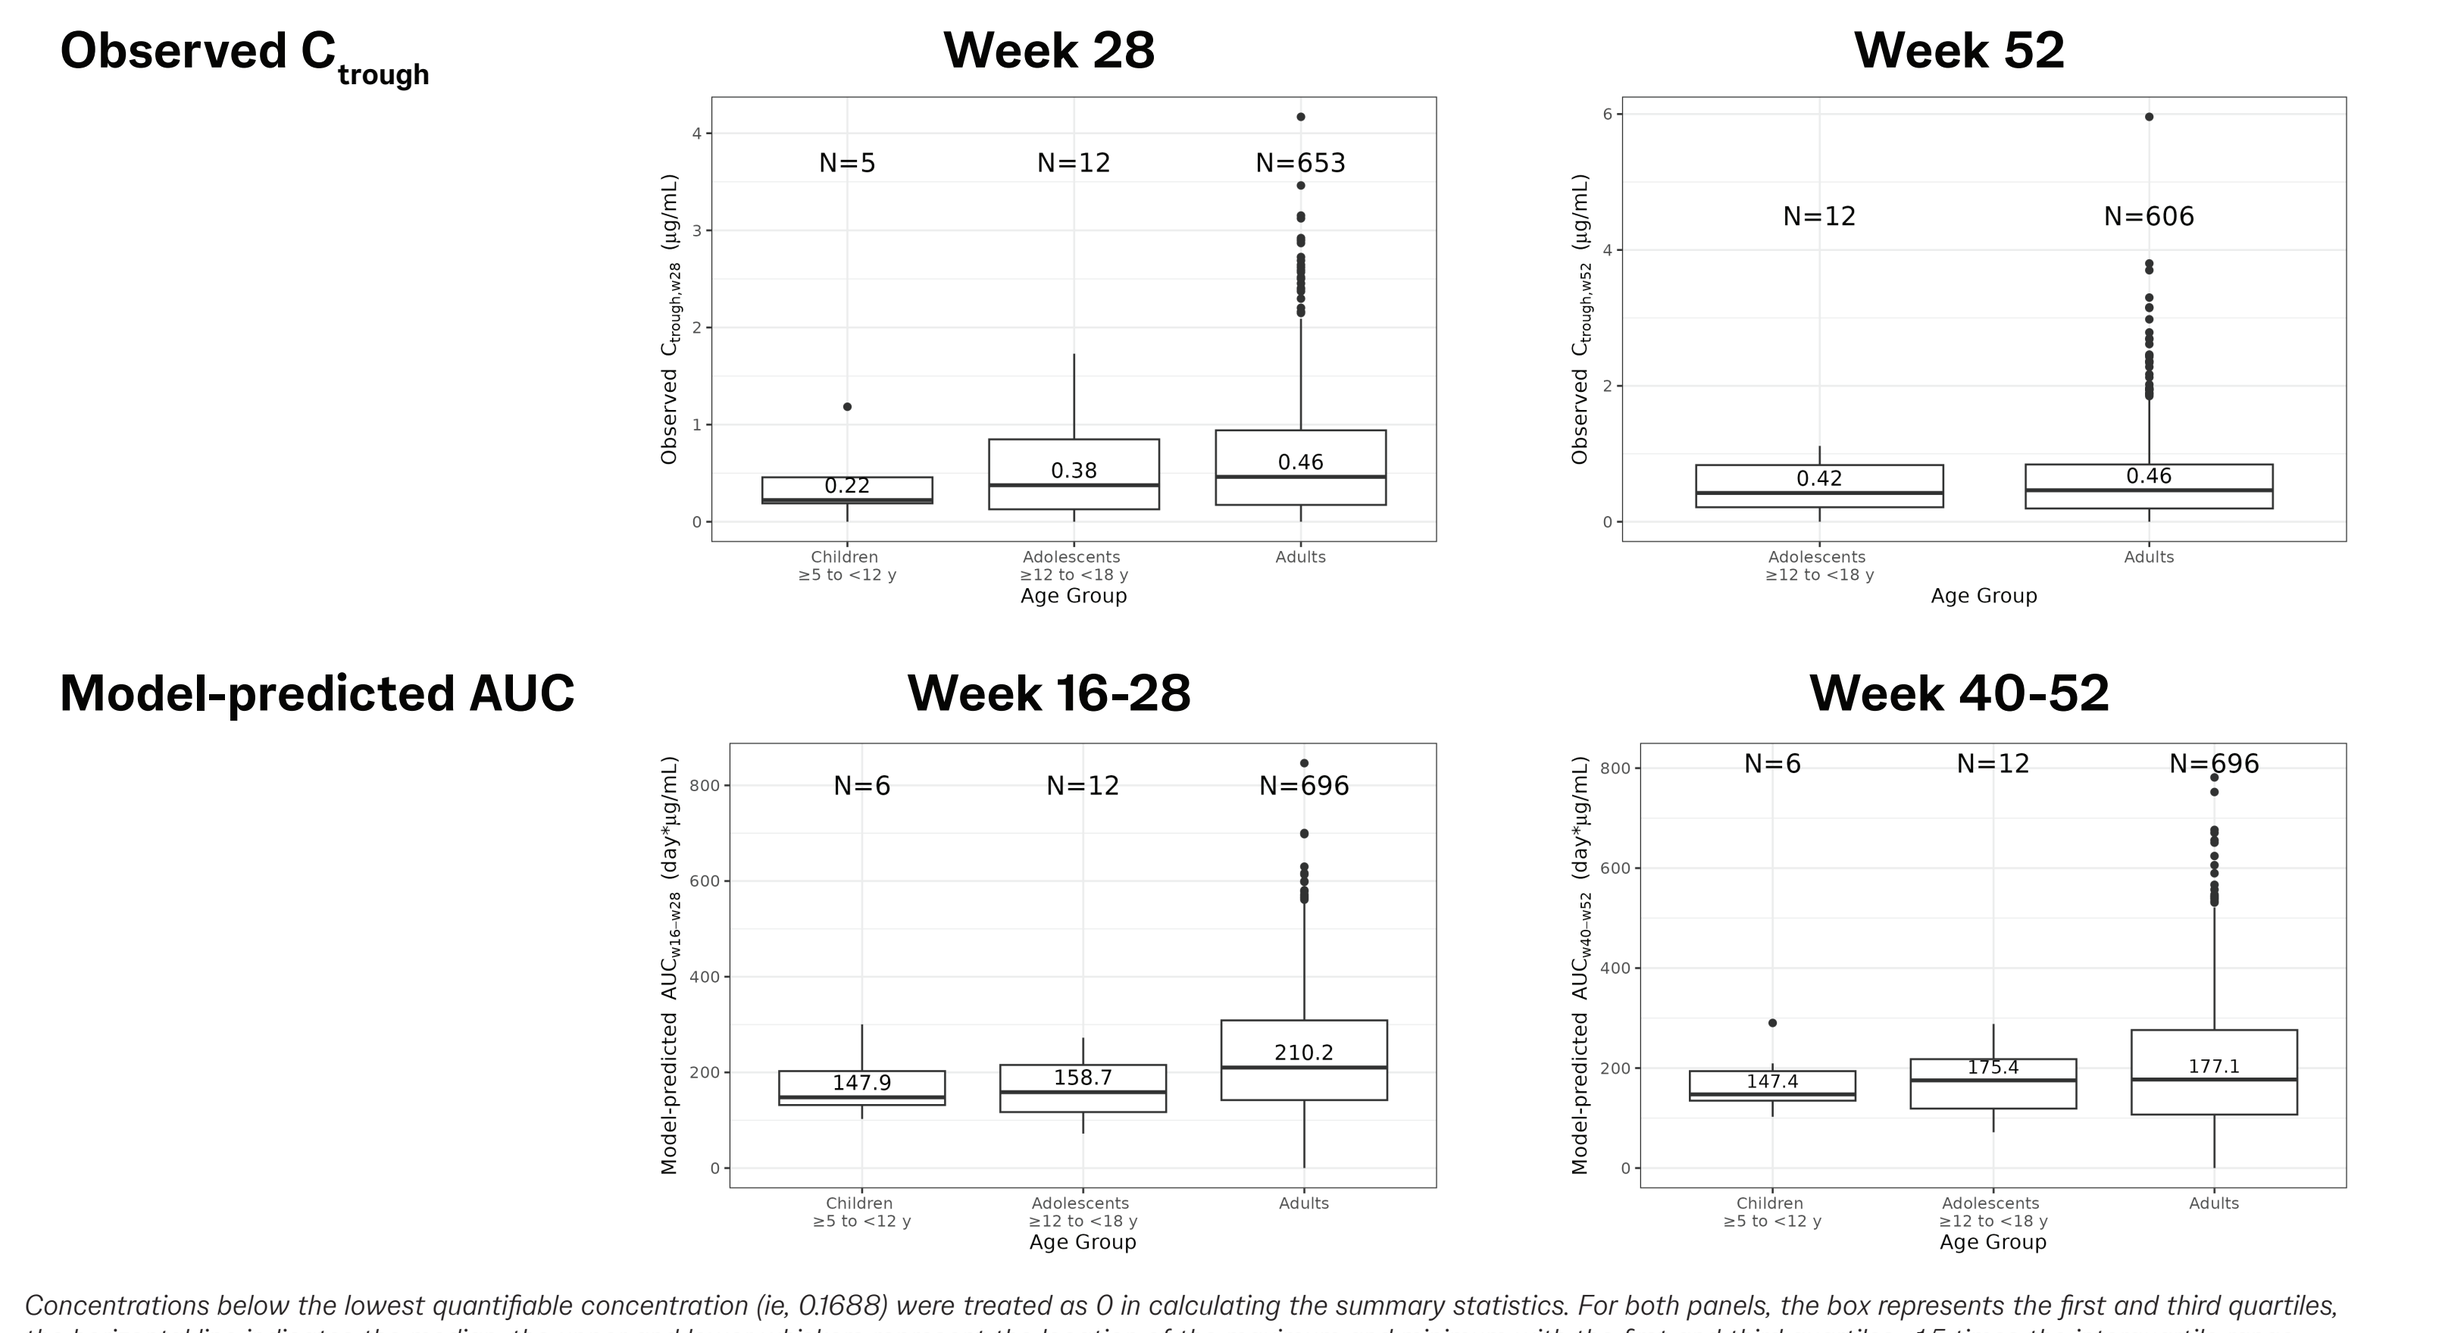
Observed C trough
Model-predicted AUC
Week 28
Week 52
Week 16-28
Week 40-52
N=5
N=12
N=653
N=606
N=6
N=696
0.22
0.38
0.46
0.42
0.46
147.9
158.7
210.2
147.4
175.4
177.1
Children ≥5 to <12 y
Adolescents ≥12 to <18 y
Adults
Age Group

Concentrations below the lowest quantifiable concentration (ie, 0.1688) were treated as 0 in calculating the summary statistics. For both panels, the box represents the first and third quartiles, the horizontal line indicates the median, the upper and lower whiskers represent the location of the maximum and minimum with the first and third quartiles ±1.5 times the interquartile range.

There was no apparent relationship between PK and JIA-ACR 30, JIA-ACR 50, or JIA-ACR 70 responses at Week 24 or 52

	UST Concentration Quartiles, μg/mL			
	0.224 to <0.538	0.538 to <1.057	1.057 to <1.850	1.850 to 2.479
Week 24, N	2	3	3	3
JIA-ACR 30, n (%)	2 (100%)	3 (100%)	3 (100%)	3 (100%)
JIA-ACR 50, n (%)	2 (100%)	3 (100%)	3 (100%)	3 (100%)
JIA-ACR 70, n (%)	2 (100%)	3 (100%)	2 (67%)	2 (67%)

	UST Concentration Quartiles, μg/mL[a]		
	0 to <0.225	0.225 to <0.661	0.661 to 1.054
Week 52, N	7	3	4
JIA-ACR 30, n (%)	4 (57%)	3 (100%)	4 (100%)
JIA-ACR 50, n (%)	4 (57%)	3 (100%)	4 (100%)
JIA-ACR 70, n (%)	4 (57%)	3 (100%)	2 (50%)

Data are based on nonresponder imputation analysis. Concentrations below the lowest quantifiable concentration (ie, 0.1688 μg/mL) were treated as zero in calculating the quartiles. [a]At Week 52, the lowest 2 quartiles were combined into one quartile. Both the minimum and first quartile of concentrations are 0.

PRESENTED AT: American College of Clinical Pharmacology; September 27–29, 2026; Bethesda, Maryland. **REFERENCES: 1.** Leu JH, et al. *Paediatr Drugs.* 2022;24:699–714. **2.** Ruperto N, et al. PReS 33rd European Paediatric Rheumatology Congress; September 16–19, 2026; Belgrade, Serbia. **ACKNOWLEDGMENTS:** Medical writing support was provided by Holly Capasso-Harris of Veristat, under the direction of the authors in accordance with Good Publication Practice guidelines (*Ann Intern Med* 2022;175:1298–1304) and was funded by Johnson & Johnson. Sponsored by Johnson & Johnson. **DISCLOSURES: TNK, YL, WZ, JHL, LB, SR, SH, VS,** and **KB** are employees of Johnson & Johnson and may own stock or stock options in Johnson & Johnson. **NR** reports honoraria for scientific consultancies or speakers' bureaus in the past 2 years from AlfaSigma, Amgen, Boehringer Ingelheim, Bristol Myers Squibb, Eli Lilly, Galapagos, Genentech Roche, Idorsia, Johnson & Johnson, Pfizer, and Takeda. **DJL** has received grant/research support from Bristol Myers Squibb, Johnson & Johnson, and Roche Laboratories and consulting fees from AstraZeneca, GlaxoSmithKline, Novartis, Pfizer (consultant and advisory board member), and United Bioscience Corporation and has served on speakers' bureaus for Novartis and Pfizer (all above are paid to institution). **AM** has received consulting fees from AbbVie, Boehringer Ingelheim, Eli Lilly, EMD Serono, Idorsia, Johnson & Johnson, Novartis, and Pfizer. **DPH, OK, HES, OB, SD, KN, CMS, AG,** and **MER** have no conflicts of interest to declare. **SC-L** reports consulting fees from Galapagos and Novartis. **BPML** reports honoraria for speakers' bureaus in the past 2 years from AstraZeneca and GlaxoSmithKline. **AB** reports personal fees from Johnson & Johnson for speaking engagements. **GF** reports honoraria for scientific consultancies or speakers' bureaus in the past 2 years from Novartis and Sobi. The Cincinnati Children's Hospital, where **HB** works as a full-time public employee, has received contributions from the following industries in the past 3 years: Bristol-Myers Squibb, Eli Lilly, GlaxoSmithKline, F. Hoffmann-La Roche, Johnson & Johnson, Novartis, and Pfizer. This funding has been reinvested for the research activities of the hospital in a fully independent manner, without any commitment to third parties. HB has received consulting fees from AbbVie, Astra Zeneca-Medimmune, Biogen, Boehringer Ingelheim, Bristol-Myers Squibb, Celgene, Eli Lilly, EMD Serono, Genzyme, GlaxoSmithKline, F. Hoffmann-La Roche, Johnson & Johnson, Merck, Novartis, R-Pharm, and Sanofi and has served on speakers' bureaus for GlaxoSmithKline, Novartis, and Roche.

This material is distributed for scientific purposes by Johnson & Johnson Innovative Medicine and is not for promotional use.