Efficacy and Safety of Guselkumab as Maintenance Therapy in Patients with Moderately to Severely Active Ulcerative Colitis: Results from the Phase 3 QUASAR Maintenance Study

David T. Rubin,[1] Jessica R. Allegretti,[2] Julián Panés,[3] Nicole Shipitofsky,[4] Shadi Yarandi,[4] Kuan-Hsiang G. Huang,[4] Matthew Germinaro,[4] Rebbecca Wilson,[4] Hongyan Zhang,[4] Hazeline Hakansson,[5] Brian G. Feagan,[6] Tadakazu Hisamatsu,[7] Gary R. Lichtenstein,[8] Brian Bressler,[9] Laurent Peyrin-Biroulet,[10] Bruce E. Sands,[11] Axel Dignass[12]

[1]University of Chicago Medicine Inflammatory Bowel Disease Center, Chicago, IL, USA; [2]Brigham and Women's Hospital Crohn's and Colitis Center, Boston, MA, USA; [3]Hospital Clínic de Barcelona, IDIBAPS, CIBERehd, Barcelona, Spain; [4]Janssen Research & Development, LLC, Spring House, PA, USA; [5]Janssen Scientific Affairs, LLC, Horsham, PA, USA; [6]Alimentiv Inc, London, ON, Canada; [7]Kyorin University, Tokyo, Japan; [8]University of Pennsylvania, Philadelphia, PA, USA; [9]University of British Columbia, Vancouver, BC, Canada; [10]Nancy University Hospital, Université de Lorraine, Nancy, France; [11]Icahn School of Medicine at Mount Sinai, New York City, NY, USA; [12]Agaplesion Markus Hospital, Frankfurt, Germany

This material is distributed for scientific purposes on Janssen Science, and is not for promotional use

Background

- Guselkumab (GUS) is a dual-acting interleukin-23 p19 (IL-23p19) subunit inhibitor that blocks IL-23 and binds to CD64, a receptor on cells that produce IL-23[1]
- Results of the QUASAR Phase 2b and Phase 3 Induction Studies of intravenous (IV) GUS in patients with moderately to severely active ulcerative colitis (UC) have been previously presented[2,3]
 - GUS subcutaneous (SC) induction is also being evaluated in inflammatory bowel disease (IBD)
- The Phase 3 QUASAR Maintenance Study (NCT04033445) is a randomized-withdrawal, double-blind, placebo-controlled study

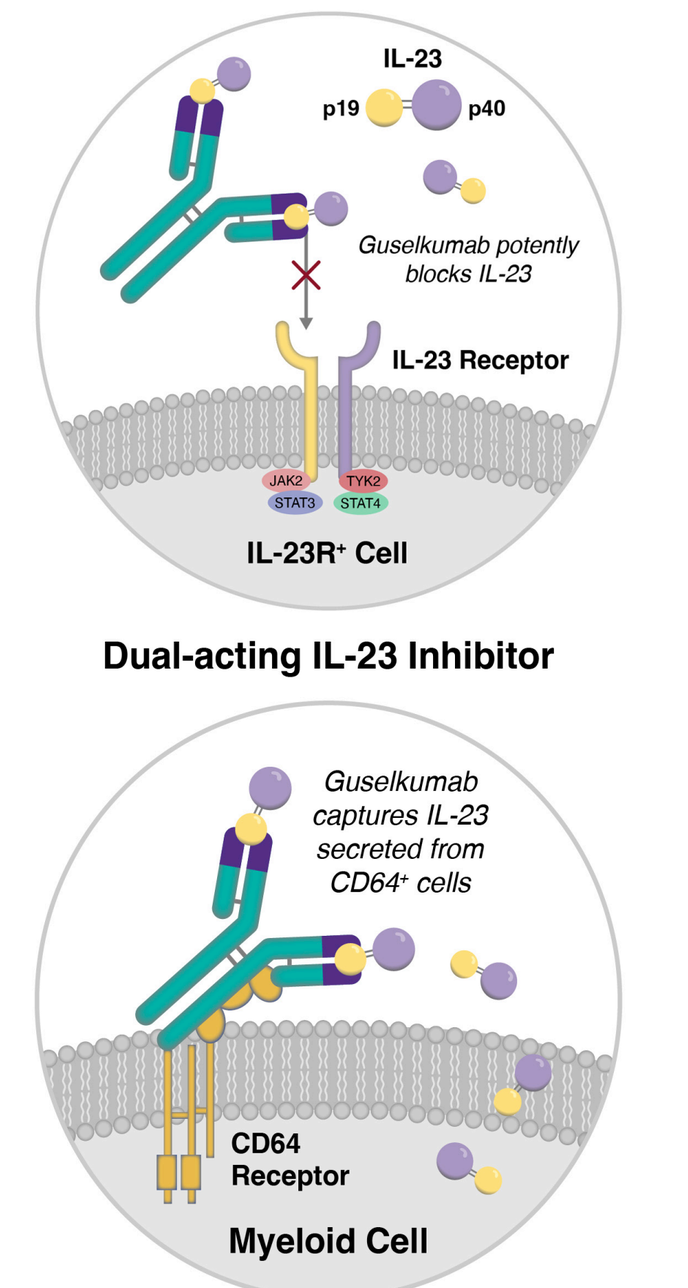
IL-23
p19
p40
Guselkumab potently blocks IL-23
IL-23 Receptor
JAK2
TYK2
STAT3
STAT4
IL-23R+ Cell
Dual-acting IL-23 Inhibitor
Guselkumab captures IL-23 secreted from CD64+ cells
CD64 Receptor
Myeloid Cell

IL=Interleukin; JAK=Janus kinase; STAT=Signal transducer and activator of transcription; TYK=Tyrosine kinase.

Objective

To report the efficacy and safety results of GUS SC maintenance treatment from the QUASAR Maintenance Study in patients with moderately to severely active UC who achieved clinical response to GUS IV induction

Methods

Target Patient Population: Adults with moderately to severely active UC, defined as induction baseline modified Mayo score of 5 to 9 with a Mayo rectal bleeding subscore ≥1 and a Mayo endoscopic subscore ≥2 based on central review

QUASAR Program Overview (N=1064, Enrolled)

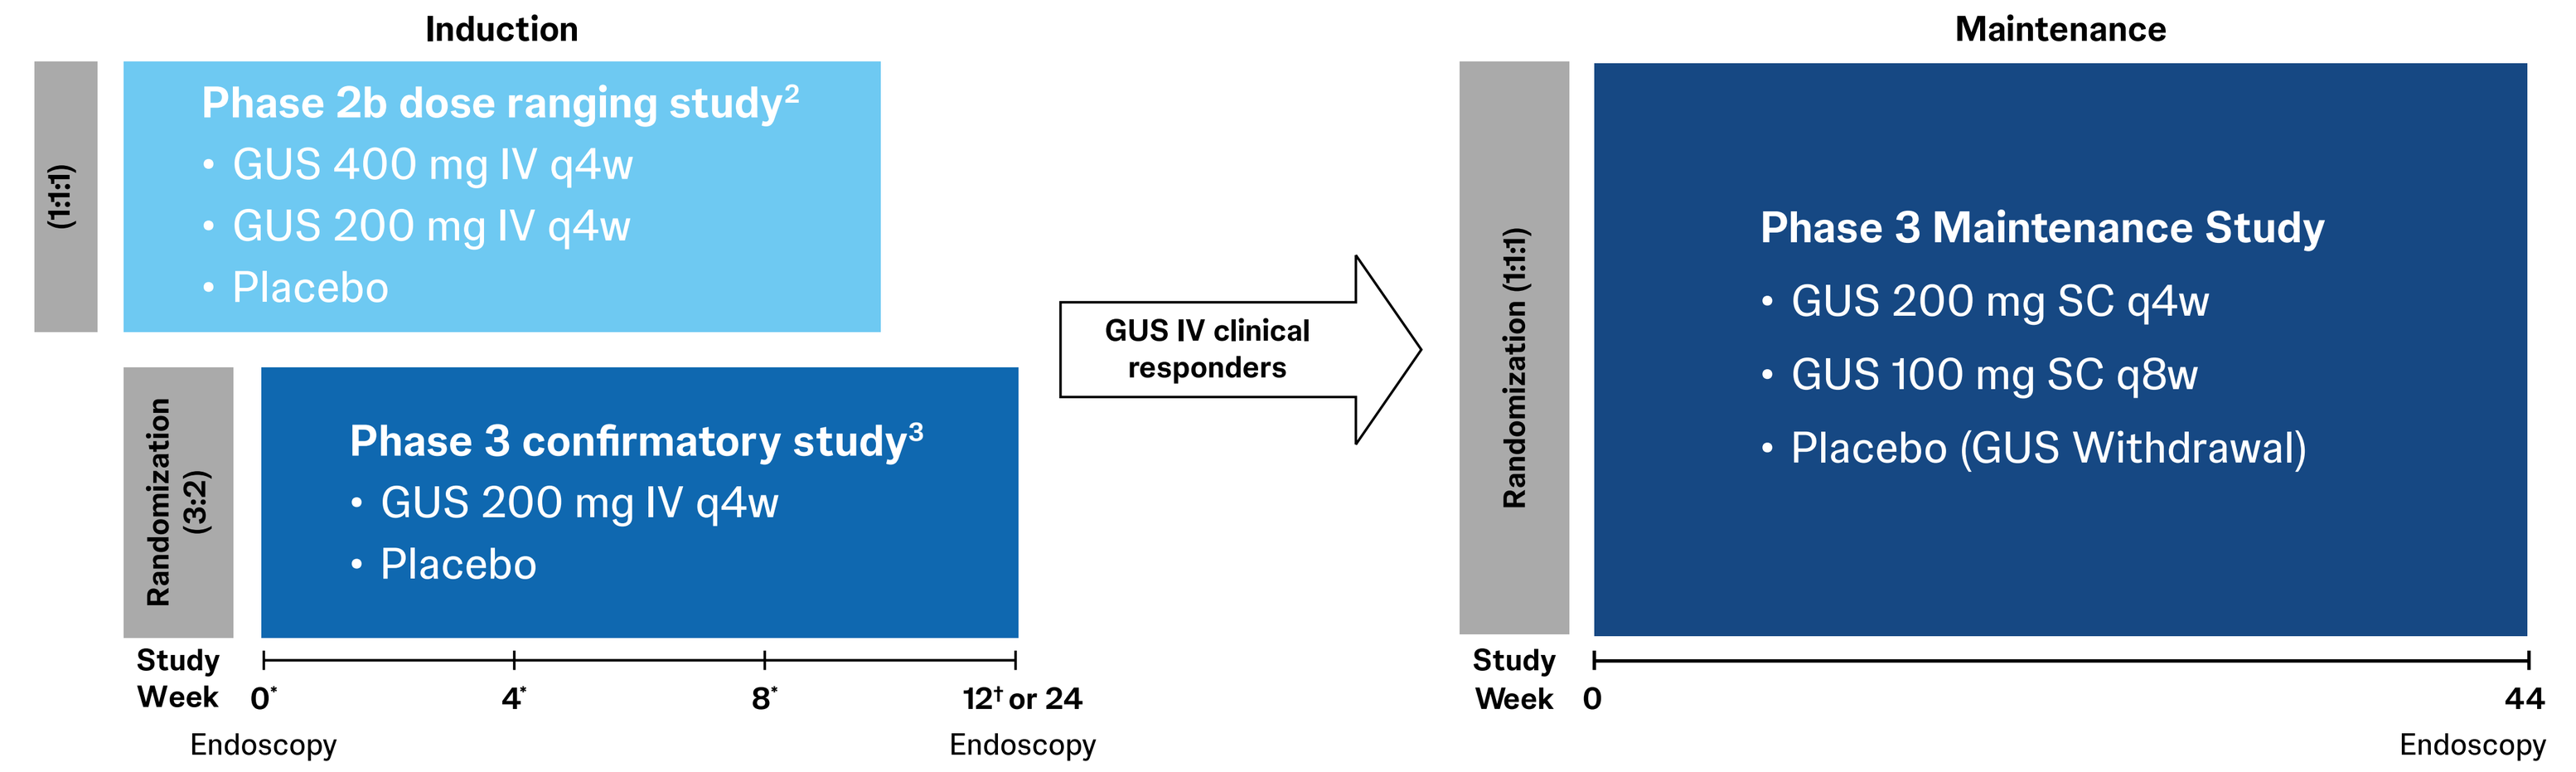
Induction
(1:1:1)
Phase 2b dose ranging study[2]
• GUS 400 mg IV q4w
• GUS 200 mg IV q4w
• Placebo
Randomization (3:2)
Phase 3 confirmatory study[3]
• GUS 200 mg IV q4w
• Placebo
Study Week 0* 4* 8* 12† or 24
Endoscopy
Endoscopy
GUS IV clinical responders
Maintenance
Randomization (1:1:1)
Phase 3 Maintenance Study
• GUS 200 mg SC q4w
• GUS 100 mg SC q8w
• Placebo (GUS Withdrawal)
Study Week 0 44
Endoscopy

All studies were double-blinded. * = Study treatment administered; † = Study treatment administered to Week 12 clinical nonresponders. GUS=Guselkumab; IV=Intravenous; q4w=Every 4 weeks; q8w=Every 8 weeks; SC=Subcutaneous; UC=Ulcerative colitis.

Primary Endpoint (Week 44)

- Clinical remission

Major Secondary Endpoints (Week 44)

Clinical
- Corticosteroid-free clinical remission
- Maintenance of clinical remission
- Maintenance of clinical response
- Symptomatic remission

Endoscopic and Histologic
- Endoscopic improvement
- Histo-endoscopic mucosal improvement
- Endoscopic remission (normalization)

Patient-reported outcomes
- Inflammatory Bowel Disease Questionnaire (IBDQ) remission
- Fatigue response

Maintenance Study Design

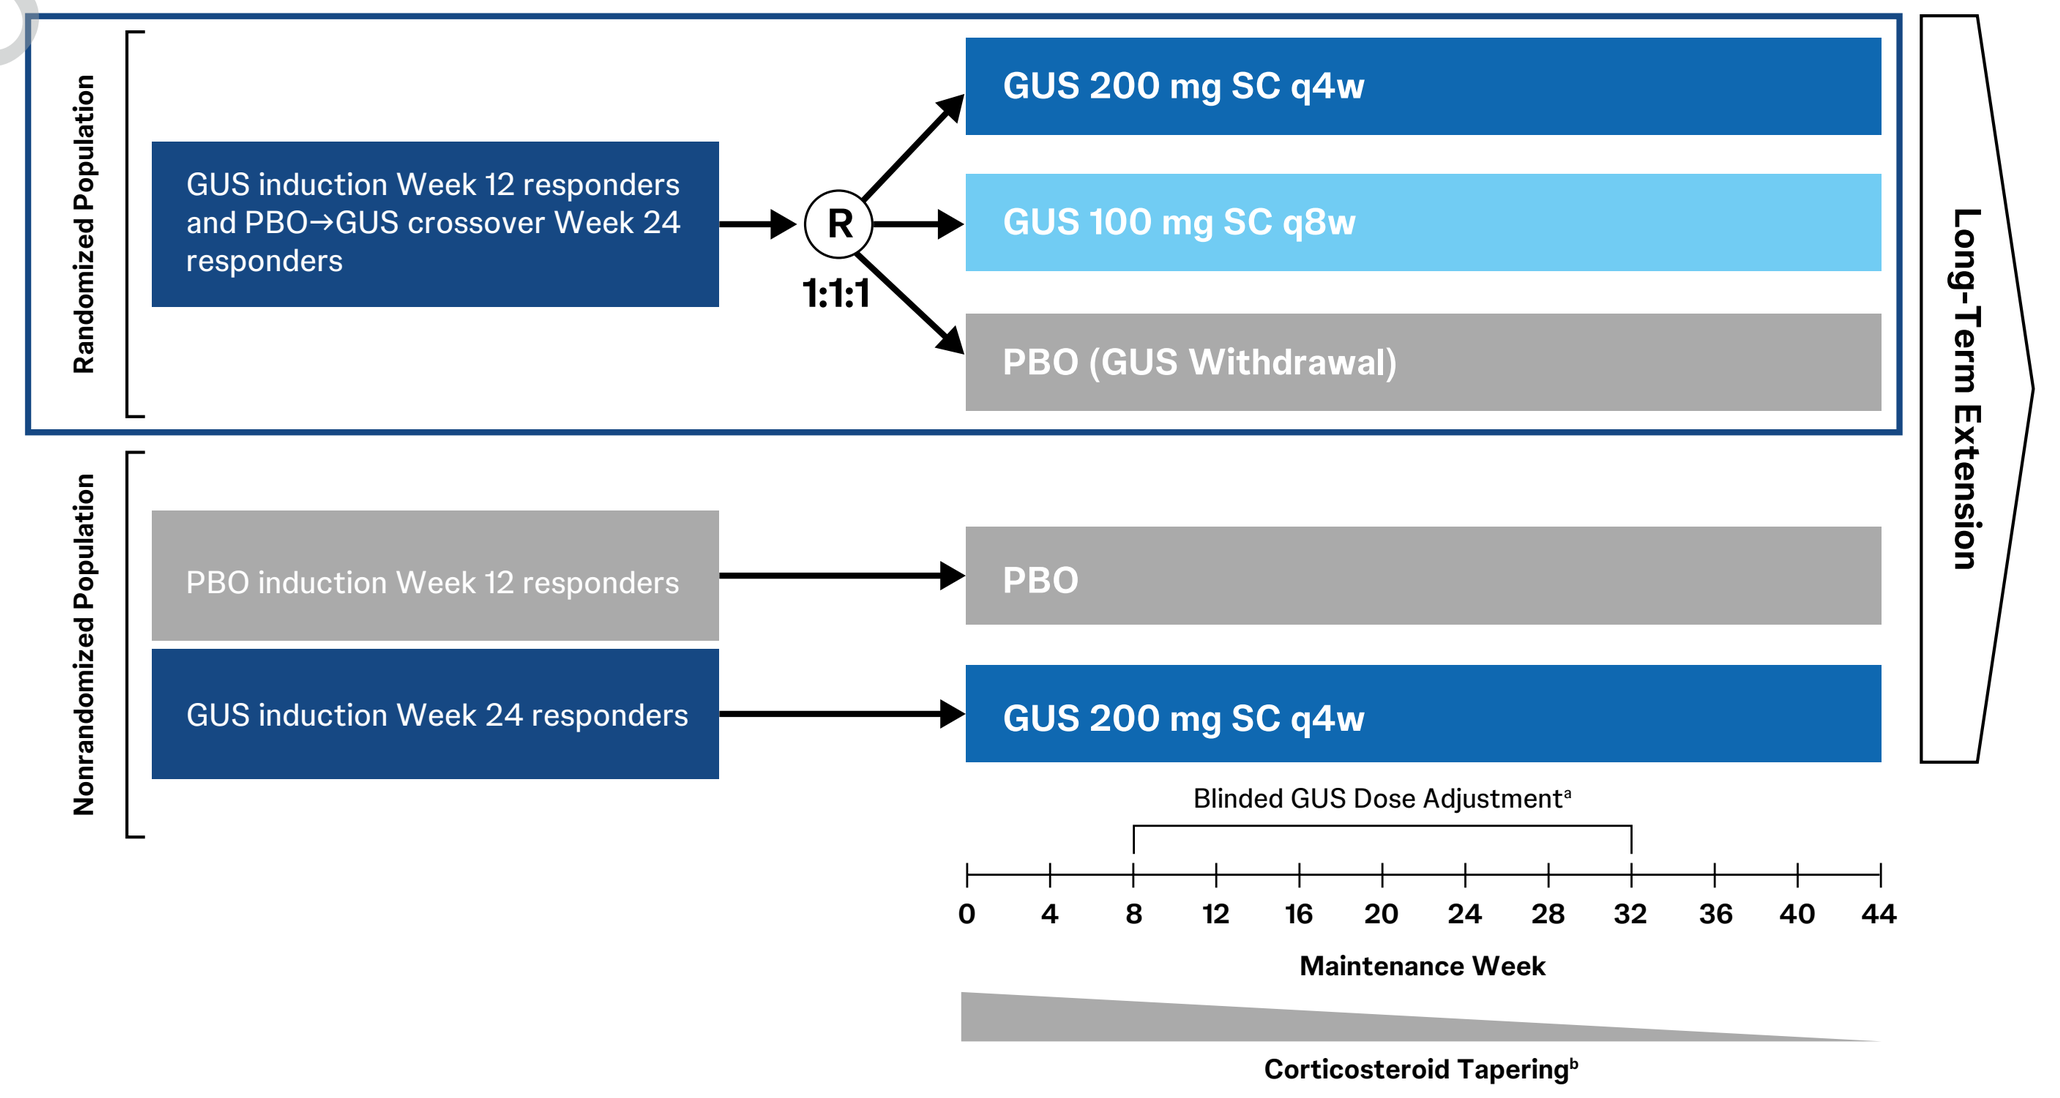
Randomized Population
GUS induction Week 12 responders and PBO→GUS crossover Week 24 responders
R
1:1:1
GUS 200 mg SC q4w
GUS 100 mg SC q8w
PBO (GUS Withdrawal)
Nonrandomized Population
PBO induction Week 12 responders
PBO
GUS induction Week 24 responders
GUS 200 mg SC q4w
Long-Term Extension
Blinded GUS Dose Adjustment[a]
0 4 8 12 16 20 24 28 32 36 40 44
Maintenance Week
Corticosteroid Tapering[b]

[a]Between Week 8 and Week 32, randomized patients meeting loss of clinical response criteria (based on the modified Mayo score and required an endoscopic assessment) were eligible for a blinded dose adjustment as follows: Placebo SC → GUS 200 mg SC q4w, GUS 100 mg SC q8w → GUS 200 mg SC q4w, GUS 200 mg SC q4w → GUS 200 mg SC q4w (sham adjustment). [b]Patients receiving corticosteroids upon maintenance entry were to begin tapering at Maintenance Week 0. Other UC-specific therapies were to be continued at stable doses unless indicated by the investigator. Tapering of corticosteroids may be paused for patients meeting clinical flare criteria. GUS=Guselkumab; PBO=Placebo; q4w=Every 4 weeks; q8w=Every 8 weeks; R=Randomization; SC=Subcutaneous; UC=Ulcerative colitis.

Key Takeaways

- **The dual-acting IL-23 inhibitor GUS was statistically superior to placebo (GUS Withdrawal) for the primary endpoint of clinical remission at Week 44. All 9 major secondary endpoints were also met, including:**
 - **Clinical and symptomatic outcomes**
 - **Endoscopic and histologic outcomes**
 - **Additional patient-reported outcomes of IBDQ remission and fatigue response**
- **Both GUS SC maintenance dose regimens were efficacious**
- **Safety results through Week 44 were consistent with the known and favorable safety profile of GUS in approved indications**

Results

Patient Characteristics at Induction Baseline

	PBO (GUS Withdrawal)	GUS 100 mg q8w	GUS 200 mg q4w	Total
Randomized full analysis set, N	190	188	190	568
Age in years, mean (SD)	41.2 (13.58)	40.3 (13.00)	40.6 (14.66)	40.7 (13.75)
Male, n (%)	109 (57.4)	102 (54.3)	100 (52.6)	311 (54.8)
UC disease duration in years, mean (SD)	7.29 (6.338)	7.78 (8.463)	8.35 (8.397)	7.81 (7.791)
Modified Mayo score (0-9), mean (SD)[a]	7.0 (1.09)	6.8 (1.15)	6.9 (1.10)	6.9 (1.12)
Modified Mayo score of 7-9 (severe), n (%)	125 (65.8%)	114 (60.6%)	124 (65.3%)	363 (63.9%)
Mayo endoscopic subscore of 3 (severe), n (%)	129 (67.9%)	125 (66.5%)	123 (64.7%)	377 (66.4%)
Extensive UC, n (%)	95 (50.0%)	79 (42.0%)	83 (43.7%)	257 (45.2%)
C-reactive protein, median in mg/L (IQR)[b]	4.2 (1.6; 8.4)	3.9 (1.4; 10.4)	3.6 (1.4; 9.1)	3.9 (1.5; 9.2)
Fecal calprotectin, median in mg/kg (IQR)[c]	1642.0 (663.0; 3498.0)	1675.0 (806.0; 3543.5)	1487.0 (603.0; 3019.0)	1605.0 (669.0; 3337.0)
Oral corticosteroid use at baseline, n (%)	77 (40.5%)	74 (39.4%)	76 (40.0%)	227 (40.0%)
Immunosuppressant use at baseline, n (%)[d]	43 (22.6%)	41 (21.8%)	42 (22.1%)	126 (22.2%)
History of inadequate response or intolerance to biologic and/or JAK inhibitor therapy, n (%)[e,f]	75 (39.5%)	77 (41.0%)	88 (46.3%)	240 (42.3%)
One biologic or JAK inhibitor, n (%)	36 (48.0%)	36 (46.8%)	52 (59.1%)	124 (51.7%)
Two or more biologics and/or JAK inhibitors, n (%)	39 (52.0%)	41 (53.2%)	36 (40.9%)	116 (48.3%)

Randomized Full Analysis Set: Randomized patients in maintenance with modified Mayo score 5-9 at induction baseline who received at least 1 Maintenance Study treatment dose. [a]Modified Mayo score: 3-component (stool frequency, rectal bleeding, and endoscopic subscores) Mayo score without the physician's global assessment. [b]Based on PBO, N=190; GUS 100 mg, N=185; GUS 200 mg, N=187; Total, N=562. [c]Based on PBO, N=175; GUS 100 mg, N=160; GUS 200 mg, N=171; Total, N=506. [d]Immunosuppressants included azathioprine, 6-mercaptopurine, and methotrexate. [e]Biologic therapy included tumor necrosis factor-α antagonists and vedolizumab. [f]JAK inhibitor therapy included facitinib. GUS=Guselkumab; IQR=Interquartile range; JAK=Janus kinase; PBO=Placebo; q4w=Every 4 weeks; q8w=Every 8 weeks; SD=Standard deviation; UC=Ulcerative colitis.

Disease Characteristics at Maintenance Baseline

	Induction Guselkumab IV Responders			
	PBO (GUS Withdrawal)	GUS 100 mg q8w	GUS 200 mg q4w	Total
Randomized full analysis set, N	190	188	190	568
Clinical remission, n (%)[a]	59 (31.1%)	66 (35.1%)	69 (36.3%)	194 (34.2%)
Endoscopic improvement, n (%)[b]	68 (35.8%)	75 (39.9%)	79 (41.6%)	222 (39.1%)
Endoscopic remission, n (%)[c]	39 (20.5%)	41 (21.8%)	47 (24.7%)	127 (22.4%)
IBDQ remission, n (%)[d]	142 (75.5%)	134 (71.3%)	128 (67.7%)	404 (71.5%)
Modified Mayo score (0-9), mean (SD)	2.5 (1.57)	2.6 (1.51)	2.5 (1.50)	2.5 (1.53)
C-reactive protein, median in mg/L (IQR)	1.5 (0.6; 4.0)	1.4 (0.4; 4.0)	1.4 (0.6; 3.4)	1.5 (0.6; 3.8)
Fecal calprotectin (mg/kg), median in mg/kg (IQR)[e]	306.5 (82.5; 1077.0)	308.0 (71.0; 1310.0)	281.0 (89.0; 1233.0)	303.5 (79.5; 1194.0)

[a]Clinical remission: A Mayo stool frequency subscore of 0 or 1 and not increased from baseline, a Mayo rectal bleeding subscore of 0, and a Mayo endoscopic subscore of 0 or 1 with no friability present. [b]Endoscopic improvement: A Mayo endoscopic subscore of 0 or 1 with no friability present. [c]Endoscopic remission (normalization): A Mayo endoscopic subscore of 0. [d]IBDQ remission: A total IBDQ score ≥ 170. Based on PBO, N=188; GUS 100 mg, N=188; GUS 200 mg, N=189; Total, N=565. [e]Based on PBO, N=188; GUS 100 mg, N=185; GUS 200 mg, N=187; Total, N=560. GUS=Guselkumab; IBDQ=Inflammatory Bowel Disease Questionnaire; IQR=Interquartile range; IV=Intravenous; PBO=Placebo; q4w=Every 4 weeks; q8w=Every 8 weeks; SD=Standard deviation.

Treatment Disposition Through Week 44

- Overall rate of treatment discontinuation was low at 12.0%
- Discontinuation due to adverse event (AE) was reported in 5.1% of all patients treated
- Most common reasons overall for discontinuation: AEs of worsening UC (3.3%) and patient withdrawal (3.3%)

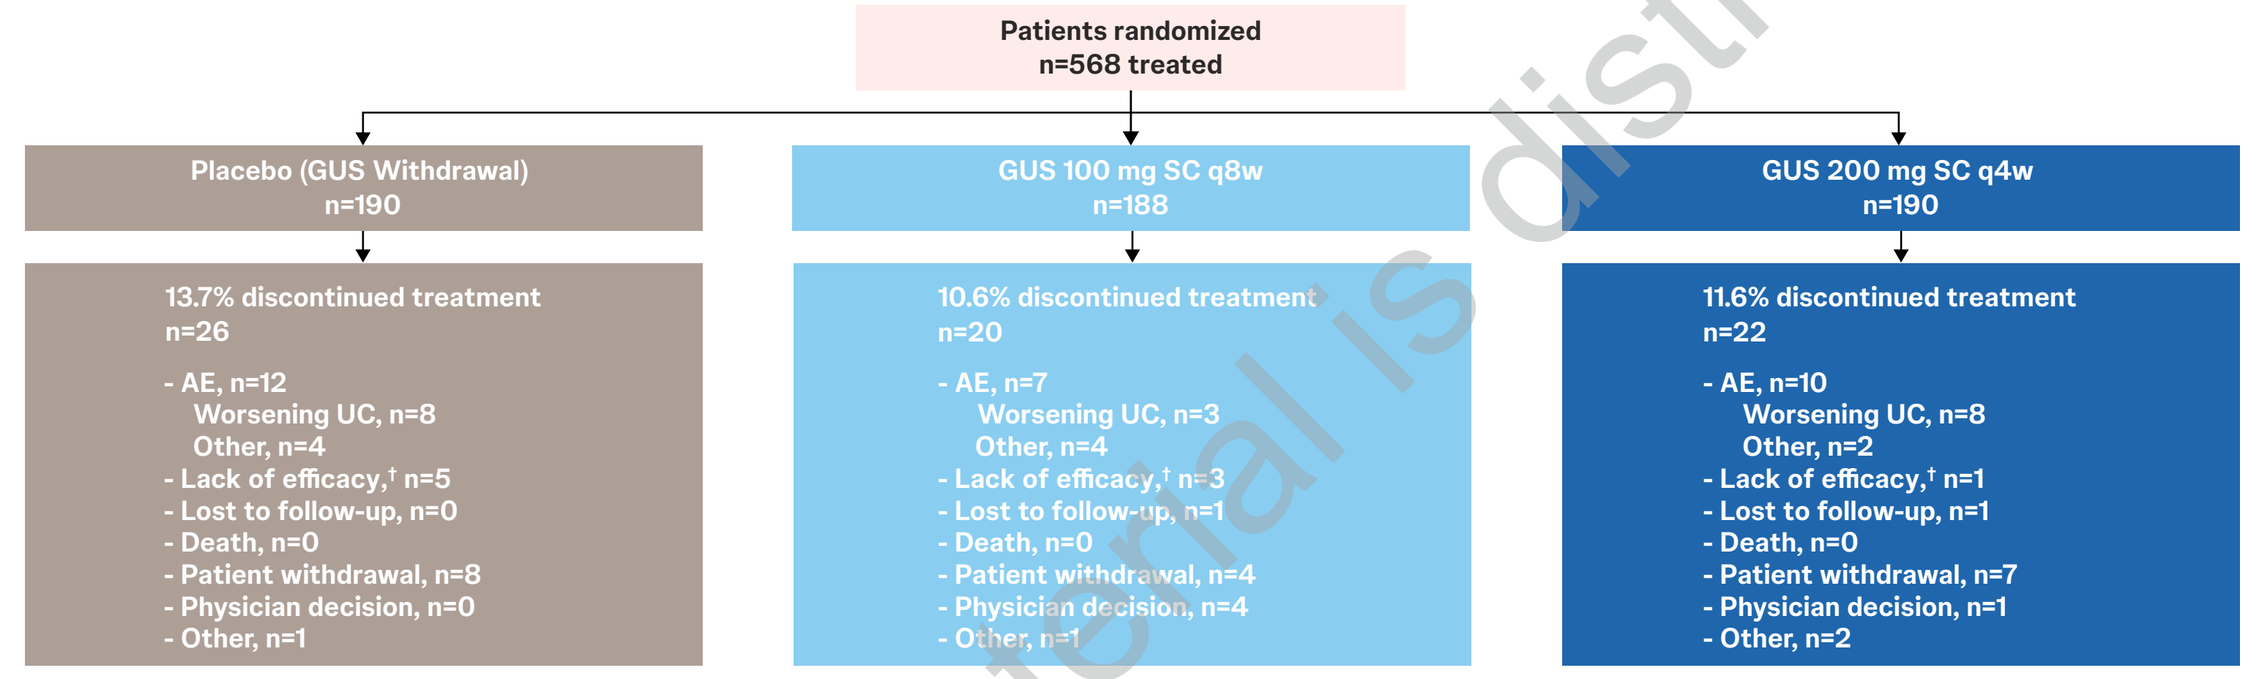
Patients randomized
n=568 treated
Placebo (GUS Withdrawal)
n=190
13.7% discontinued treatment
n=26
- AE, n=12
Worsening UC, n=8
Other, n=4
- Lack of efficacy,† n=5
- Lost to follow-up, n=0
- Death, n=0
- Patient withdrawal, n=8
- Physician decision, n=0
- Other, n=1
GUS 100 mg SC q8w
n=188
10.6% discontinued treatment
n=20
- AE, n=7
Worsening UC, n=3
Other, n=4
- Lack of efficacy,† n=3
- Lost to follow-up, n=1
- Death, n=0
- Patient withdrawal, n=4
- Physician decision, n=4
- Other, n=1
GUS 200 mg SC q4w
n=190
11.6% discontinued treatment
n=22
- AE, n=10
Worsening UC, n=8
Other, n=2
- Lack of efficacy,† n=1
- Lost to follow-up, n=1
- Death, n=0
- Patient withdrawal, n=7
- Physician decision, n=1
- Other, n=2

Randomized Full Analysis Set. As a result of an urgent site closure due to regional crisis in Russia and Ukraine, 3 patients did not have available treatment disposition status at Week 44. †As determined by the investigator. AE=Adverse event; GUS=Guselkumab; q4w=Every 4 weeks; q8w=Every 8 weeks; SC=Subcutaneous; UC=Ulcerative colitis.

Primary Endpoint: Clinical Remission at Week 44

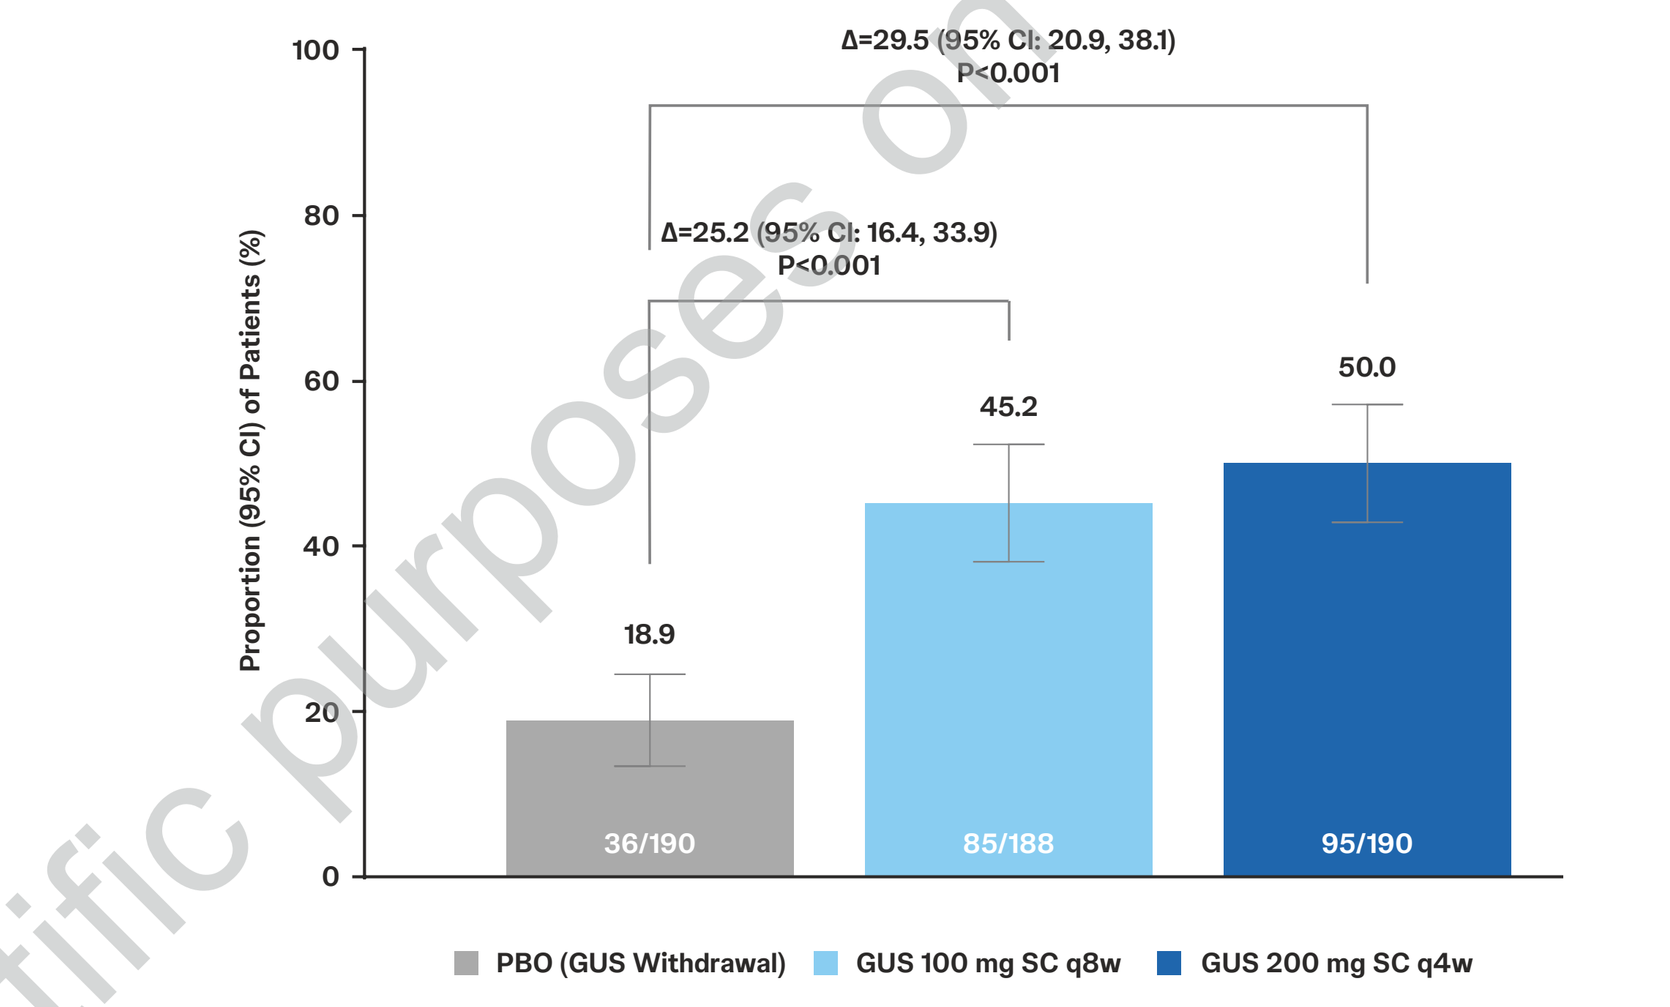
Δ=29.5 (95% CI: 20.9, 38.1)
P<0.001
Δ=25.2 (95% CI: 16.4, 33.9)
P<0.001
Proportion (95% CI) of Patients (%)
100
80
60
40
20
0
18.9
45.2
50.0
36/190
85/188
95/190
PBO (GUS Withdrawal)
GUS 100 mg SC q8w
GUS 200 mg SC q4w

69% of GUS-treated patients in clinical remission were also in endoscopic remission (MES=0)

Randomized Full Analysis Set. **Clinical remission:** A Mayo stool frequency subscore of 0 or 1 and not increased from baseline, a Mayo rectal bleeding subscore of 0, and MES of 0 or 1 with no friability present. CI=Confidence interval; GUS=Guselkumab; MES=Mayo endoscopic subscore; PBO=Placebo; q4w=Every 4 weeks; q8w=Every 8 weeks; SC=Subcutaneous.

Major Secondary Clinical Endpoints At Week 44

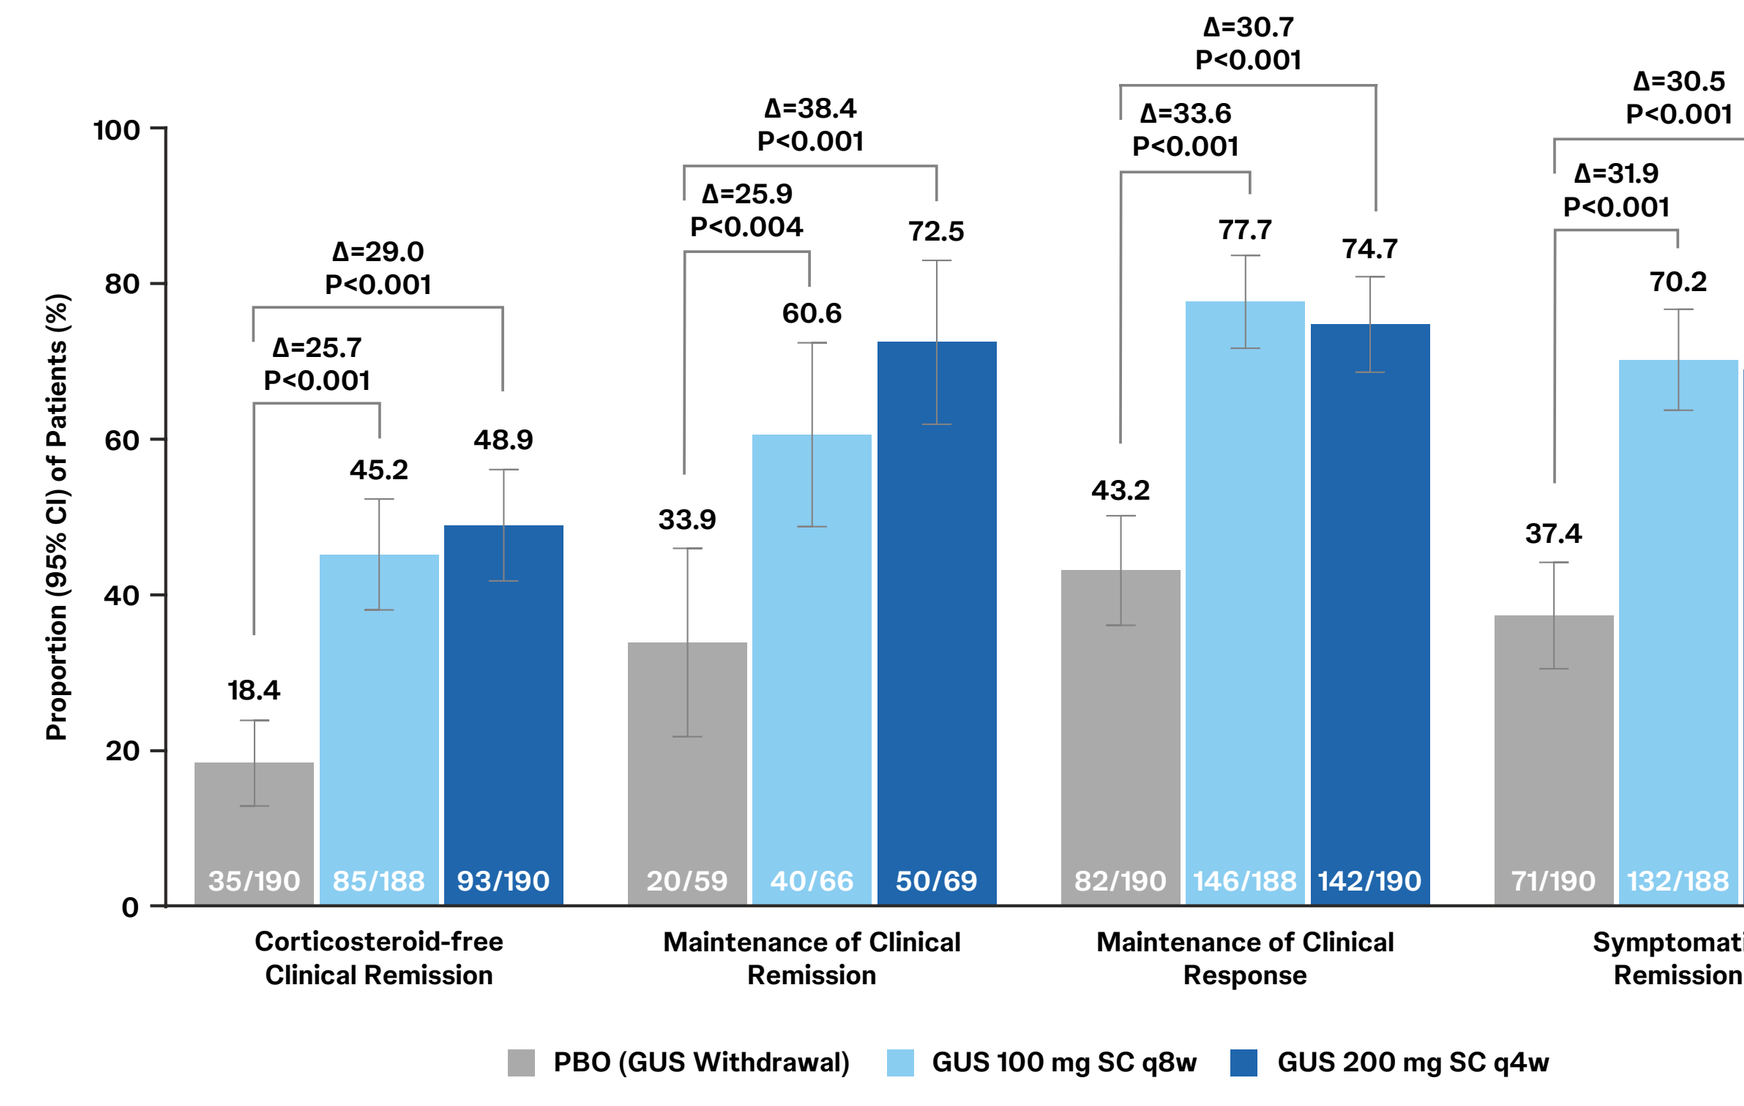
Proportion (95% CI) of Patients (%)
Δ=29.0
P<0.001
Δ=25.7
P<0.001
18.4
45.2
48.9
35/190
85/188
93/190
Corticosteroid-free Clinical Remission
Δ=38.4
P<0.001
Δ=25.9
P<0.004
33.9
60.6
72.5
20/59
40/66
50/69
Maintenance of Clinical Remission
Δ=30.7
P<0.001
Δ=33.6
P<0.001
43.2
77.7
74.7
82/190
146/188
142/190
Maintenance of Clinical Response
Δ=30.5
P<0.001
Δ=31.9
P<0.001
37.4
70.2
68.9
71/190
132/188
131/190
Symptomatic Remission
PBO (GUS Withdrawal)
GUS 100 mg SC q8w
GUS 200 mg SC q4w

Randomized Full Analysis Set. **Corticosteroid-free clinical remission:** Not requiring any treatment with corticosteroids for ≥8 weeks prior to Week 44 and meeting the criteria for clinical remission at Week 44. **Maintenance of clinical remission:** Clinical remission at Week 44 among patients in clinical remission at maintenance baseline. **Maintenance of clinical response:** Clinical response at Week 44 among patients in clinical response at maintenance baseline. **Symptomatic remission:** A stool frequency subscore of 0 or 1 and not increased from induction baseline, and a rectal bleeding subscore of 0. CI=Confidence interval; GUS=Guselkumab; PBO=Placebo; q4w=Every 4 weeks; q8w=Every 8 weeks; SC=Subcutaneous.

Symptomatic Remission Through Week 44

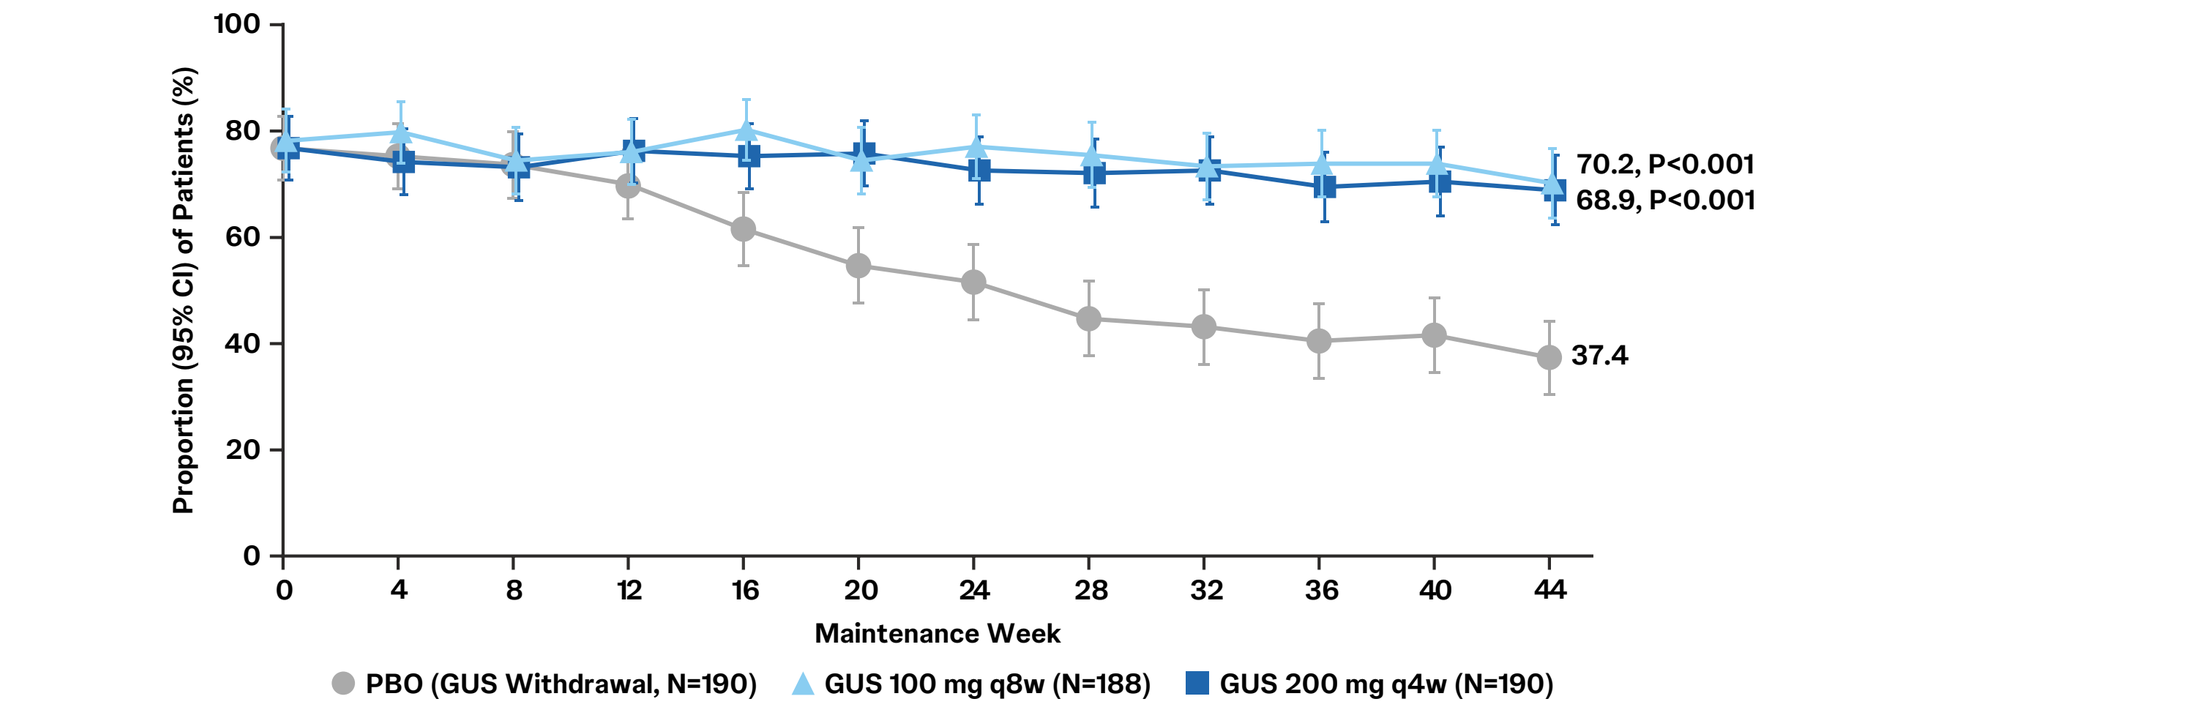
Proportion (95% CI) of Patients (%)
100
80
60
40
20
0
0 4 8 12 16 20 24 28 32 36 40 44
Maintenance Week
70.2, P<0.001
68.9, P<0.001
37.4
PBO (GUS Withdrawal, N=190)
GUS 100 mg q8w (N=188)
GUS 200 mg q4w (N=190)

Randomized Full Analysis Set. **Symptomatic remission:** A stool frequency subscore of 0 or 1 and not increased from baseline, and a rectal bleeding subscore of 0. CI=Confidence interval; GUS=Guselkumab; PBO=Placebo; q4w=Every 4 weeks; q8w=Every 8 weeks.

Major Secondary Endoscopic and Histologic Endpoints at Week 44

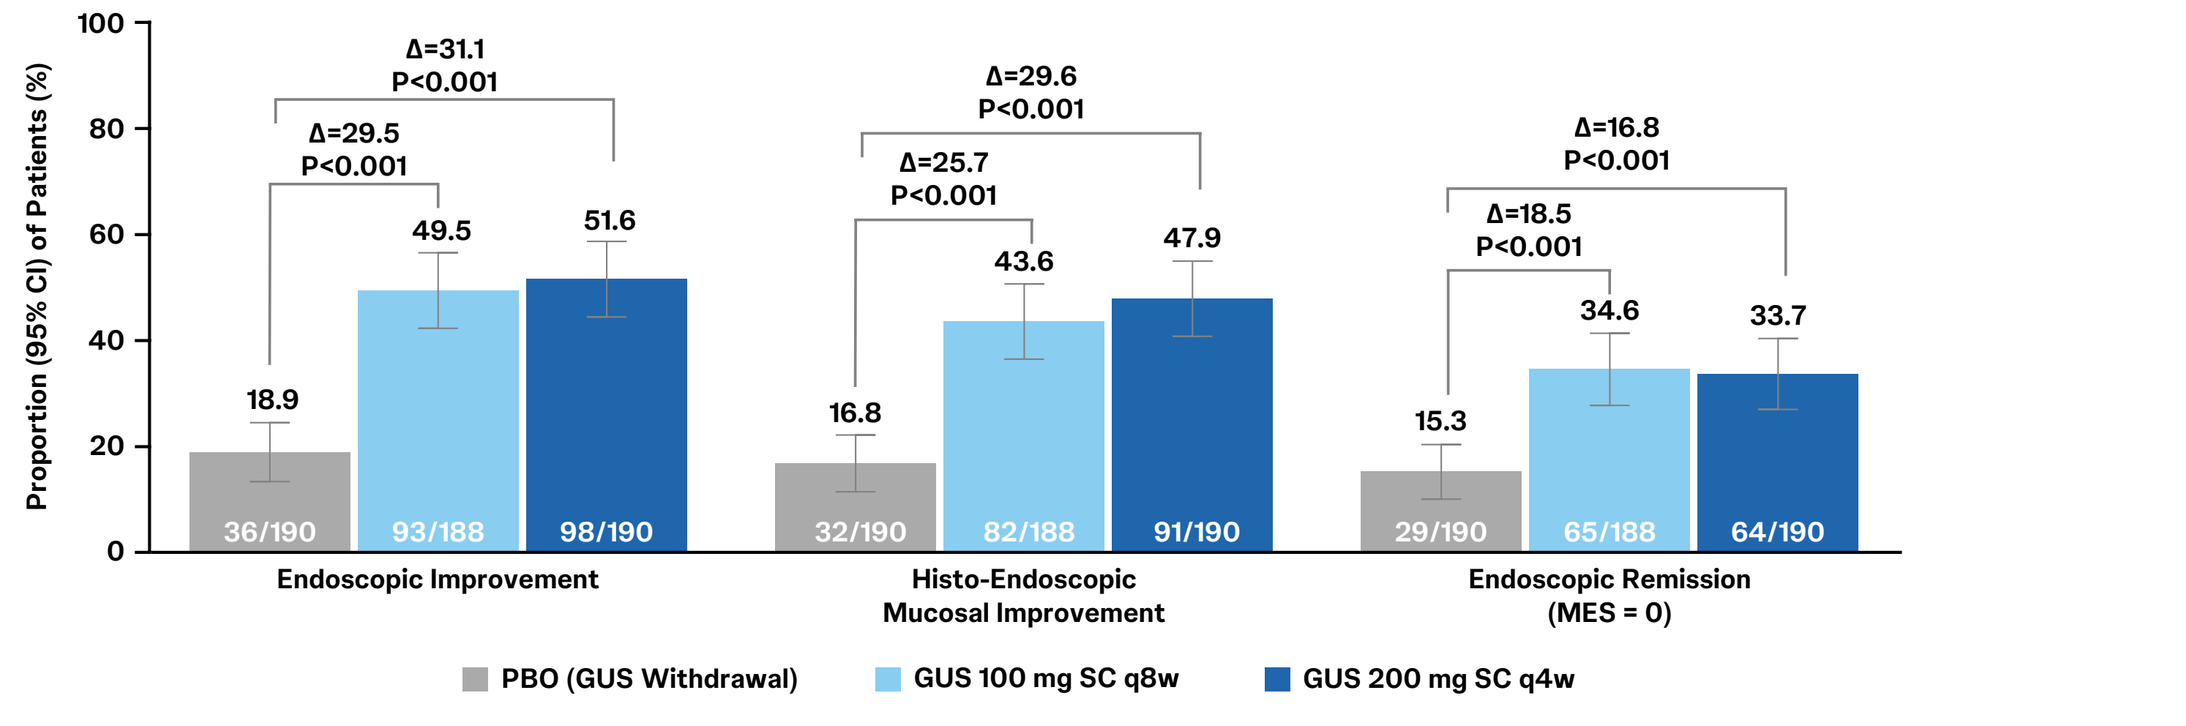
Proportion (95% CI) of Patients (%)
Δ=31.1
P<0.001
Δ=29.5
P<0.001
18.9
49.5
51.6
36/190
93/188
98/190
Endoscopic Improvement
Δ=29.6
P<0.001
Δ=25.7
P<0.001
16.8
43.6
47.9
32/190
82/188
91/190
Histo-Endoscopic Mucosal Improvement
Δ=16.8
P<0.001
Δ=18.5
P<0.001
15.3
34.6
33.7
29/190
65/188
64/190
Endoscopic Remission (MES = 0)
PBO (GUS Withdrawal)
GUS 100 mg SC q8w
GUS 200 mg SC q4w

Randomized Full Analysis Set. **Endoscopic improvement:** MES = 0 or 1 with no friability present. **Histo-endoscopic mucosal improvement:** Achieving a combination of histologic improvement (neutrophil infiltration in <5% of crypts, no crypt destruction, and no erosions, ulcerations or granulation tissue according to the Geboes grading system) and endoscopic improvement. **Endoscopic remission (normalization):** MES = 0. CI=Confidence interval; GUS=Guselkumab; MES=Mayo endoscopic subscore; PBO=Placebo; q4w=Every 4 weeks; q8w=Every 8 weeks; SC=Subcutaneous.

Major Secondary Patient-Reported Outcome Endpoints at Week 44

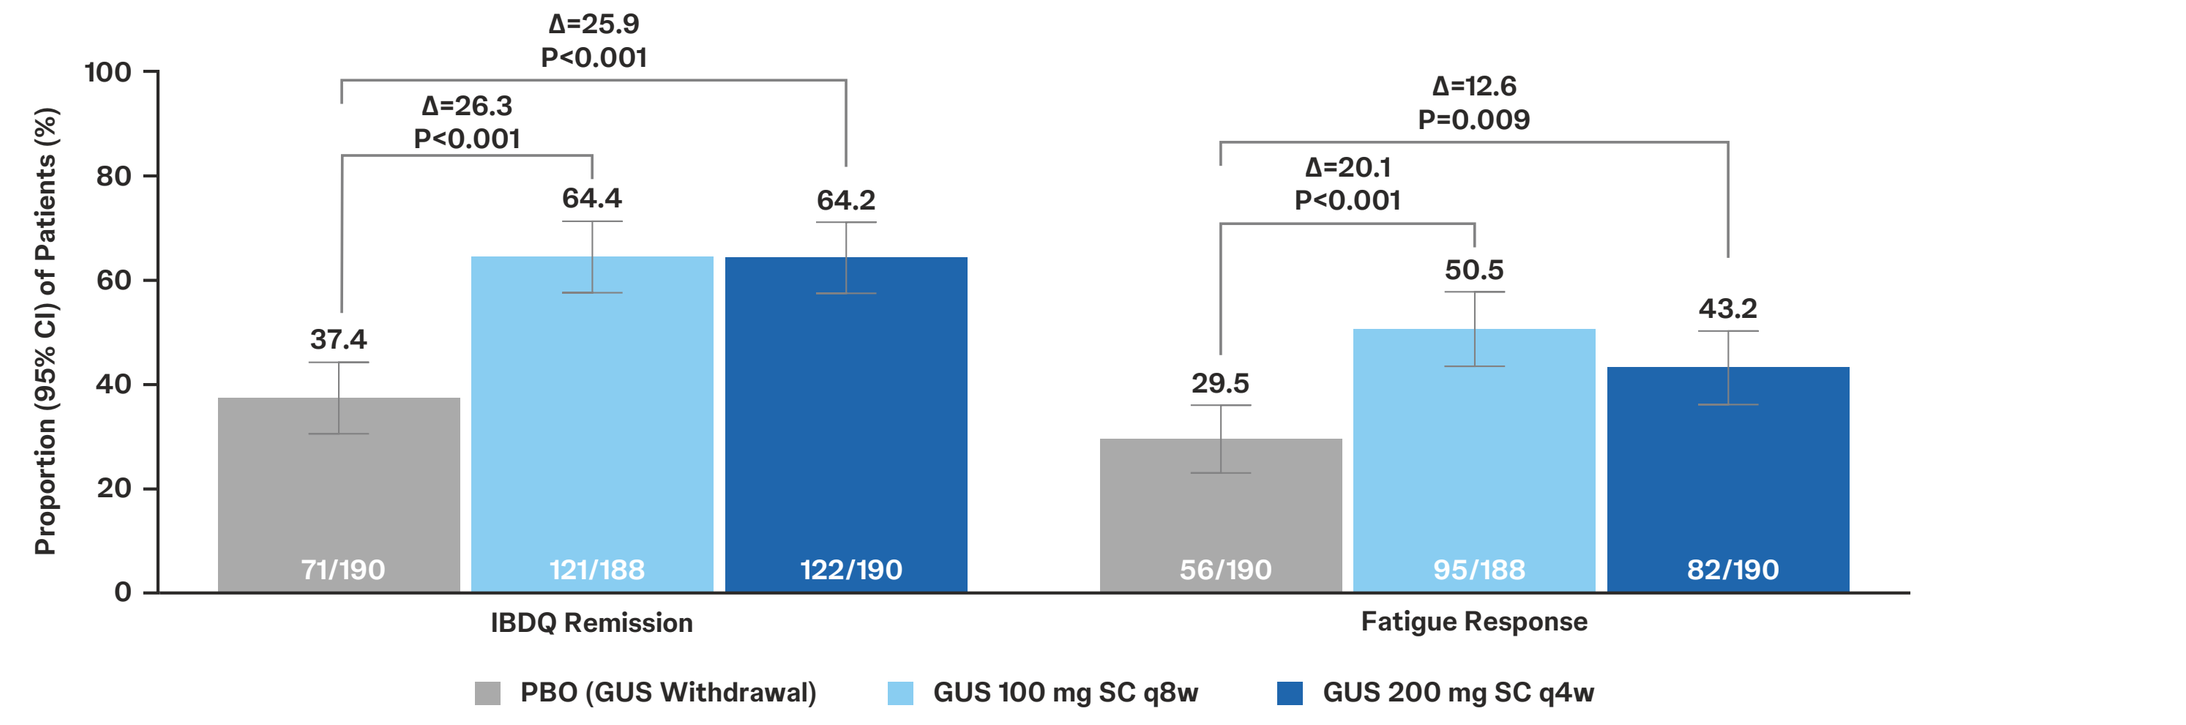
Proportion (95% CI) of Patients (%)
Δ=25.9
P<0.001
Δ=26.3
P<0.001
37.4
64.4
64.2
71/190
121/188
122/190
IBDQ Remission
Δ=12.6
P=0.009
Δ=20.1
P<0.001
29.5
50.5
43.2
56/190
95/188
82/190
Fatigue Response
PBO (GUS Withdrawal)
GUS 100 mg SC q8w
GUS 200 mg SC q4w

Randomized Full Analysis Set. **IBDQ remission:** A total IBDQ score ≥ 170. **Fatigue response:** A ≥ 7-point improvement from induction baseline in the PROMIS Fatigue Short Form 7a. CI=Confidence interval; GUS=Guselkumab; IBDQ=Inflammatory Bowel Disease Questionnaire; PBO=Placebo; PROMIS=Patient-Reported Outcomes Measurement Information System; q4w=Every 4 weeks; q8w=Every 8 weeks; SC=Subcutaneous.

Summary of Adverse Events Through Week 44

		Randomized GUS	
	Randomized PBO (GUS withdrawal)	100 mg q8w	200 mg q4w
Randomized safety analysis set, N	192	186	190
Average duration of follow-up, weeks	34.0	40.5	39.2
Average exposure, number of administrations	8.2	9.9	9.6
Deaths	0	0	0
Patients with 1 or more, n (%):			
AEs	131 (68.2%)	120 (64.5%)	133 (70.0%)
Serious AEs	1 (0.5%)	5 (2.7%)	12 (6.3%)
AEs leading to discontinuation	13 (6.8%)	7 (3.8%)	5 (2.6%)
Infections	63 (32.8%)	59 (31.7%)	59 (31.1%)
Serious infections	0	1 (0.5%)	2 (1.1%)

Most common AEs among GUS-treated patients:	COVID-19	UC	Arthralgia
	14.1% PBO 11.2% GUS Combined	29.7% PBO 11.2% GUS Combined	6.8% PBO 6.1% GUS Combined

Includes only patients with modified Mayo score 5-9 at induction baseline who were randomized in the Maintenance Study and data up to the time of dose adjustment for patients who had a dose adjustment. AE=Adverse event; COVID-19=Coronavirus disease 2019; GUS=Guselkumab; PBO=Placebo; q4w=Every 4 weeks; q8w=Every 8 weeks; UC=Ulcerative colitis.

Summary of Targeted Adverse Events Through Week 44

		Randomized GUS	
	Randomized PBO (GUS withdrawal)	100 mg q8w	200 mg q4w
Randomized safety analysis set, N	192	186	190
Average duration of follow-up, weeks	34.0	40.5	39.2
Average duration of treatment, weeks	29.2	32.6	34.9
Patients with 1 or more, n (%):			
Active tuberculosis	0	0	0
Malignancies	4 (2.1%)	0	1 (0.5%)
Anaphylactic reactions	0	0	0
Serum sickness reactions	0	0	0
Opportunistic infections	0	0	0
Major adverse cardiovascular events	0	0	1 (0.5%)
Venous thromboembolism	0	0	1 (0.5%)
Clinically important hepatic disorders[a]	0	0	0

Includes only patients with modified Mayo score 5-9 at induction baseline who were randomized in the Maintenance Study and data up to the time of dose adjustment for patients who had a dose adjustment. Patients were counted only once for any given event. [a]Defined as hepatic disorder adverse events reported as serious adverse events or adverse events leading to discontinuation of study agent. GUS=Guselkumab; PBO=Placebo; q4w=Every 4 weeks; q8w=Every 8 weeks.

PRESENTED AT: Congress of Clinical Rheumatology-West (CCR-W); San Diego, CA, USA, September 26-29, 2024.

REFERENCES: 1. Atreya R, Abreu MT, Krueger JG, et al. *J Crohns Colitis.* 2024;18(suppl):S470. **2.** Peyrin-Biroulet L, Allegretti JR, Rubin DT, et al. *Gastroenterology.* 2023 Dec;165(6):1443-1457. **3.** Allegretti JR, Peyrin-Biroulet L, Feagan BG, et al. *Gastroenterology.* 2023; 164(suppl):S1572.

ACKNOWLEDGEMENTS: The authors thank the patients, investigators, and study personnel who made the QUASAR study possible. Under the direction of the authors and in accordance with Good Publication Practices, Nichole Orlowsky of Janssen Scientific Affairs, LLC provided writing and editorial assistance. Layout design and reformatting for this encore presentation was provided by Samita Warang of Siro Clinpharm Pvt Ltd, Thane, Maharashtra, India. Sponsored by Janssen Research & Development, LLC, a Johnson & Johnson Company.

DISCLOSURES: DTR reports research funding from Takeda; consultant to AbbVie, Altrubio, Allergan, Inc., Arena Pharmaceuticals, Aslan Pharmaceuticals, Athos Therapeutics, Bellatrix Pharmaceuticals, Boehringer Ingelheim, Ltd., Bristol Myers Squibb, Celgene Corp/Syneos, Connect BioPharma, GalenPharma/Atlantica, Genentech/Roche, InDex Pharmaceuticals, Ironwood Pharmaceuticals, Iterative Scopes, Janssen Pharmaceuticals, Eli Lilly, Materia Prima, Pfizer, Prometheus Biosciences, Reistone, Takeda, and Techlab, Inc; co-founder of Cornerstones Health, Inc. **JRA** is a consultant for Finch Therapeutics, Artugen, Pfizer, Takeda, Janssen, and Iterative Scopes; receives research support from Merck; consultant and speaker for BMS. **JP** reports grants from Abbvie and Pfizer; consulting fees/honorarium from Abbvie, Arena, Athos, Boehringer Ingelheim, Celgene, Celltrion, Genentech - Roche, GSK, Janssen, Mirum, Morphic, Origo, Pandion, Pfizer, Progenity, Revolo, Takeda, Theravance, and Wassermann, TiGenix, Arena, Neste, Pfizer, and Progenity; support for travel to meetings from Abbvie and Takeda, during the conduct of the study; payment for lectures including service on speakers bureaus from Abbott, Takeda, Janssen, and Pfizer; payment for development of educational presentations from Abbott, Roche, Pfizer, and Janssen. **NS, SY, KGH, MG, RW, HZ, HH** are employees of Johnson & Johnson and own company stock/stock options. **BGF** received grant/research support from AbbVie Inc., Amgen Inc., AstraZeneca/MedImmune Ltd., Atlantic Pharmaceuticals Ltd., Boehringer-Ingelheim, Celgene Corporation, Celltech, Genentech Inc/Hoffmann-La Roche Ltd., Gilead Sciences Inc., GlaxoSmithKline (GSK), Janssen Research & Development LLC., Pfizer Inc., Receptos Inc./Celgene International, Sanofi, Santarus Inc., Takeda Development Center Americas Inc., Tillotts Pharma AG, and UCB; a consultant for Abbott/AbbVie, AdMIRx Inc., Akebia Therapeutics, Allergan, Amgen, Applied Molecular Transport Inc., Aptevo Therapeutics, Asta Pharma, Astra Zeneca, Atlantic Pharma, Avir Pharma, Biogen Idec, BioMx Israel, Boehringer-Ingelheim, Boston Pharmaceuticals, Bristol-Myers Squibb, Calypso Biotech, Celgene, Elan/Biogen, EnGene, Ferring Pharma, Roche/Genentech, Galapagos, Galen/Atlantica, GiCare Pharma, Gilead, Gossamer Pharma, GSK, Inception IBD Inc, Intact Therapeutics, JnJ/Janssen, Kyowa Kakko Kirin Co Ltd., Lexicon, Lilly, Lycera BioTech, Merck, Mesoblast Pharma, Millennium, Nestles, Nextbiotix, Novonordisk, ParImmune, Parvus Therapeutics Inc., Pfizer, Prometheus Therapeutics and Diagnostics, Progenity, Protagonist, Qu Biologics, Rebiotix, Receptos, Salix Pharma, Shire, Sienna Biologics, Sigmoid Pharma, Sterna Biologicals, Synergy Pharma Inc., Takeda, Teva Pharma, TiGenix, Tillotts, UCB Pharma, Vertex Pharma, Vivelix Pharma, VHsquared Ltd., and Zyngenia; a member of the speakers bureau for Abbott/AbbVie, JnJ/Janssen, Lilly, Takeda, Tillotts, UCB Pharma; a member of the scientific advisory board for Abbott/AbbVie, Allergan, Amgen, Astra Zeneca, Atlantic Pharma, Avaxia Biologics Inc., Boehringer-Ingelheim, Bristol-Myers Squibb, Celgene, Centocor Inc., Elan/Biogen, Galapagos, Genentech/Roche, JnJ/Janssen, Merck, Nestles, Novartis, Novonordisk, Pfizer, Prometheus Laboratories, Protagonist, Salix Pharma, Sterna Biologicals, Takeda, Teva, TiGenix, Tillotts Pharma AG, UCB Pharma; Senior Scientific Officer – Robarts Clinical Trials Inc. **TH** received grant support from AbbVie, Daiichi-Sankyo, EA Pharma Co, Ltd. JIMRO, Mitsubishi Tanabe Pharma Corporation, Mochida Pharmacuetical Co., Ltd., Nippon Kayaku Co., Ltd., Pfizer Inc., and Takeda Pharmaceutical Co., Ltd.; consulting fees from EA Pharma Co, Ltd., and Janssen Research & Development, LLC.; lecture fees from AbbVie, EA Pharma Co, Ltd, Mitsubishi Tanabe Pharma Corporation, and Takeda Pharmaceutical Co., Ltd. **GRL** discloses relationships with Abbvie (Consultant), Allergan (Consultant), American College of Gastroenterology (Honorarium for Associate Editor of American Journal of Gastroenterology), American Gastroenterological Association (CME), American Regent (Consultant, Honorarium [CME Program]), Celgene (Research, Consultant), Chemed (CME), Eli Lilly (Consultant, Data Safety Monitoring Board), Endo Pharmaceuticals (Consultant), Ferring (Consultant), Gastroenterology and Hepatology (Gastro-Hep Communication, Editor-Honorarium), Gilead (Consultant), IMEDEX (CME), Ironwood (CME), Janssen/ Janssen Orthobiotech (Consultant, Research, Funding to University of PA [IBD Fellow Education]), MedEd Consultants (Consultant), Merck (Consulting, Honorarium [CME Program]), Morphic Therapeutics (Consultant), Pfizer Pharmaceuticals (Consultant, Funding to University of PA [IBD Fellow Education]), Professional Communications, Inc. (Royalty for writing Textbook), Prometheus Laboratories, Inc (Consultant), Romark (Consultant, Honorarium for CME), Sandoz (Consultant), Salix Pharmaceuticals/Valeant (Consultant), Shire Pharmaceuticals (Consultant), SLACK, Inc (Book Royalty), Springer Science and Business Media (Editor [Honorarium]), Takeda (Consultant, Funding to University of PA [IBD Fellow Education], Research), University of Kentucky (CME), UCB (Consultant, Research), Up-To-Date (Author [Honorarium]), Vindico (CME), and Virgo (Consultant [Stock options]). **BB** is an advisor/speaker for Abbvie, BMS, Ferring, Janssen, Merck, Novartis, Organon, Pfizer, Sandoz, Takeda; advisor for Alimentiv, Allergan, AMT, Bausch Health, BMS, Celgene, Celltrion Healthcare, Eupraxia, Fresenius Kabi, Genentech, Gilead, Iterative Scopes, Jamp Pharma, Merck Amgen, Microbiome Insights, Mylan, Pendopharm, Protagonist, and Viatris; received research support from Abbvie, Amgen, BI, BMS, Genentech, GSK, Janssen, Merck, and Qu Biologic; and reports stock options for Qu Biologic. **LPB** reports personal fees from AbbVie, Allergan, Alma, Amgen, Applied Molecular Transport, Arena, Biogen, Boehringer Ingelheim, Bristol-Myers Squibb, Celgene, Celltrion, Enterome, Enthera, Ferring, Fresenius, Genentech, Gilead, Hikma, Index Pharmaceuticals, Janssen, Lilly, MSD, Mylan, Nestlé, Norgine, Oppilan Pharma, OSE Immunotherapeutics, Pfizer, Pharmacosmos, Roche, Samsung Bioepis, Sandoz, Sterna, Sublimity Therapeutics, Takeda, Tillots, and Vifor; grants from AbbVie, MSD, and Takeda; stock options from CTMA. **BES** reports consulting fees from Abbvie, Alimentiv, Amgen, Arena Pharmaceuticals, Artugen Therapeutics, Astra Zeneca, Boehringer Ingelheim, Boston Pharmaceuticals, Calibr, Celgene, Celltrion, ClostraBio, Equillium, Enthera, Evommune, Fresenius Kabi, Galapagos, Genentech (Roche), Gilead Sciences, GlaxoSmithKline, Gossamer Bio, Index Pharmaceuticals, Innovation Pharmaceuticals, Inotrem, Kaleido, Kallyope, Merck, Morphic Therapeutics, MRM Health, Progenity, Prometheus Biosciences, Prometheus Laboratories, Protagonist Therapeutics, Q32 Bio, Sun Pharma, Surrozen, Target RWE,Teva, TLL Pharmaceutical, and Ventyx Biosciences; consulting and speaking fees from Abivax; consulting and speaking fees and other support from Bristol Myers Squibb, Janssen, Pfizer, and Takeda; research grants and consulting fees from Theravance Biopharma; stock options from Ventyx Biopharma. **AD** reports fees for participation in clinical trials, review activities such as data monitoring boards, statistical analysis and end point committees from Abivax, AbbVie, Arena Pharmaceuticals, Bristol Myers Squibb/Celgene, Dr Falk Foundation, Galapagos, Gilead, Janssen, and Pfizer; consultancy fees from AbbVie, Amgen, Arena Pharmaceuticals, Biogen, Boehringer Ingelheim, Bristol Myers Squibb/Celgene, Celltrion, Dr Falk Foundation, Ferring Pharmaceuticals, Fresenius Kabi, Galapagos, Janssen, Lilly, MSD, Pfizer, Pharmacosmos, Roche/Genentech, Sandoz/Hexal, Takeda, Tillotts, and Vifor Pharma; payment from lectures including service on speakers bureaus from AbbVie, Biogen, CED Service GmbH, Celltrion, Falk Foundation, Ferring, Galapagos, Gilead, High5MD, Janssen, Materia Prima, MedToday, MSD, Pfizer, Streamed-Up, Takeda, Tillotts, and Vifor Pharma; payment for manuscript preparation from Falk Foundation, Takeda, Thieme, and UniMed Verlag.

Previously presented at DDW; Washington, D.C., USA; May 18-21, 2024 and FSR 2024; Orlando, FL, USA; July 11-14, 2024 and RhAPP 2024; Nashville, TN, USA; September 26-28, 2024.