PXXXX

On-label persistence through 24 months in patients with psoriatic arthritis using guselkumab or subcutaneous interleukin-17A inhibitors

Philip J Mease,[1,2] Jessica Walsh,[3,4] Timothy P Fitzgerald,[5] Soumya D Chakravarty,[5,6] Bruno Emond,[7] Carmine Rossi,[7] Samuel Schwartzbein,[7] Kana Yokoji,[7] Yuxi Wang,[7] Patrick Lefebvre,[7] Dominic Pilon,[7] Linda Hou,[5] Shikha Singla,[8] Joseph F Merola[9]

[1]Rheumatology Research, Providence Swedish Medical Center, Seattle, WA, USA; [2]University of Washington School of Medicine, Seattle, WA, USA; [3]Salt Lake City Veterans Affairs Health, Salt Lake City, UT, USA; [4]University of Utah Health, Salt Lake City, UT, USA; [5]Janssen Scientific Affairs, LLC, a Johnson & Johnson company, Horsham, PA, USA; [6]Drexel University College of Medicine, Philadelphia, PA, USA; [7]Analysis Group, Inc., Montreal, QC, Canada; [8]Medical College of Wisconsin, Milwaukee, WI, USA; [9]Department of Dermatology, and Department of Medicine, Division of Rheumatology, UT Southwestern Medical Center, Dallas, TX, USA

This material is distributed for scientific purposes on Janssen Science, and is not for promotional use

Background

- Guselkumab (GUS), a fully human IL-23 p19-subunit inhibitor, was approved by the US Food and Drug Administration (FDA) for the treatment of active psoriatic arthritis (PsA) in July 2020
 - FDA-approved dosing regimen[1] (on-label): GUS 100 mg at week 0, week 4, then every 8 weeks
- A previous claims-based analysis compared on-label persistence for patients (pts) with PsA initiating treatment with on-label GUS or their first subcutaneous (SC) interleukin-17A inhibitor (IL-17Ai)[2]
 - Pts receiving GUS were significantly (~2x) more likely to remain persistent through 12 months
- Long-term claims data comparing GUS and SC IL-17Ai persistence beyond 12 months provide additional real-world evidence about treatment persistence in routine clinical care that may differ from stringently controlled clinical trial settings

Objectives

This study utilized health plan claims data to compare treatment persistence through 24 months between pts with active PsA newly initiating an on-label GUS dosing regimen and those starting an initial SC IL-17Ai

Methods

IQVIA™ Health Plan Claims Data (1/1/2011-6/30/2023)[a] Study design[b-d]

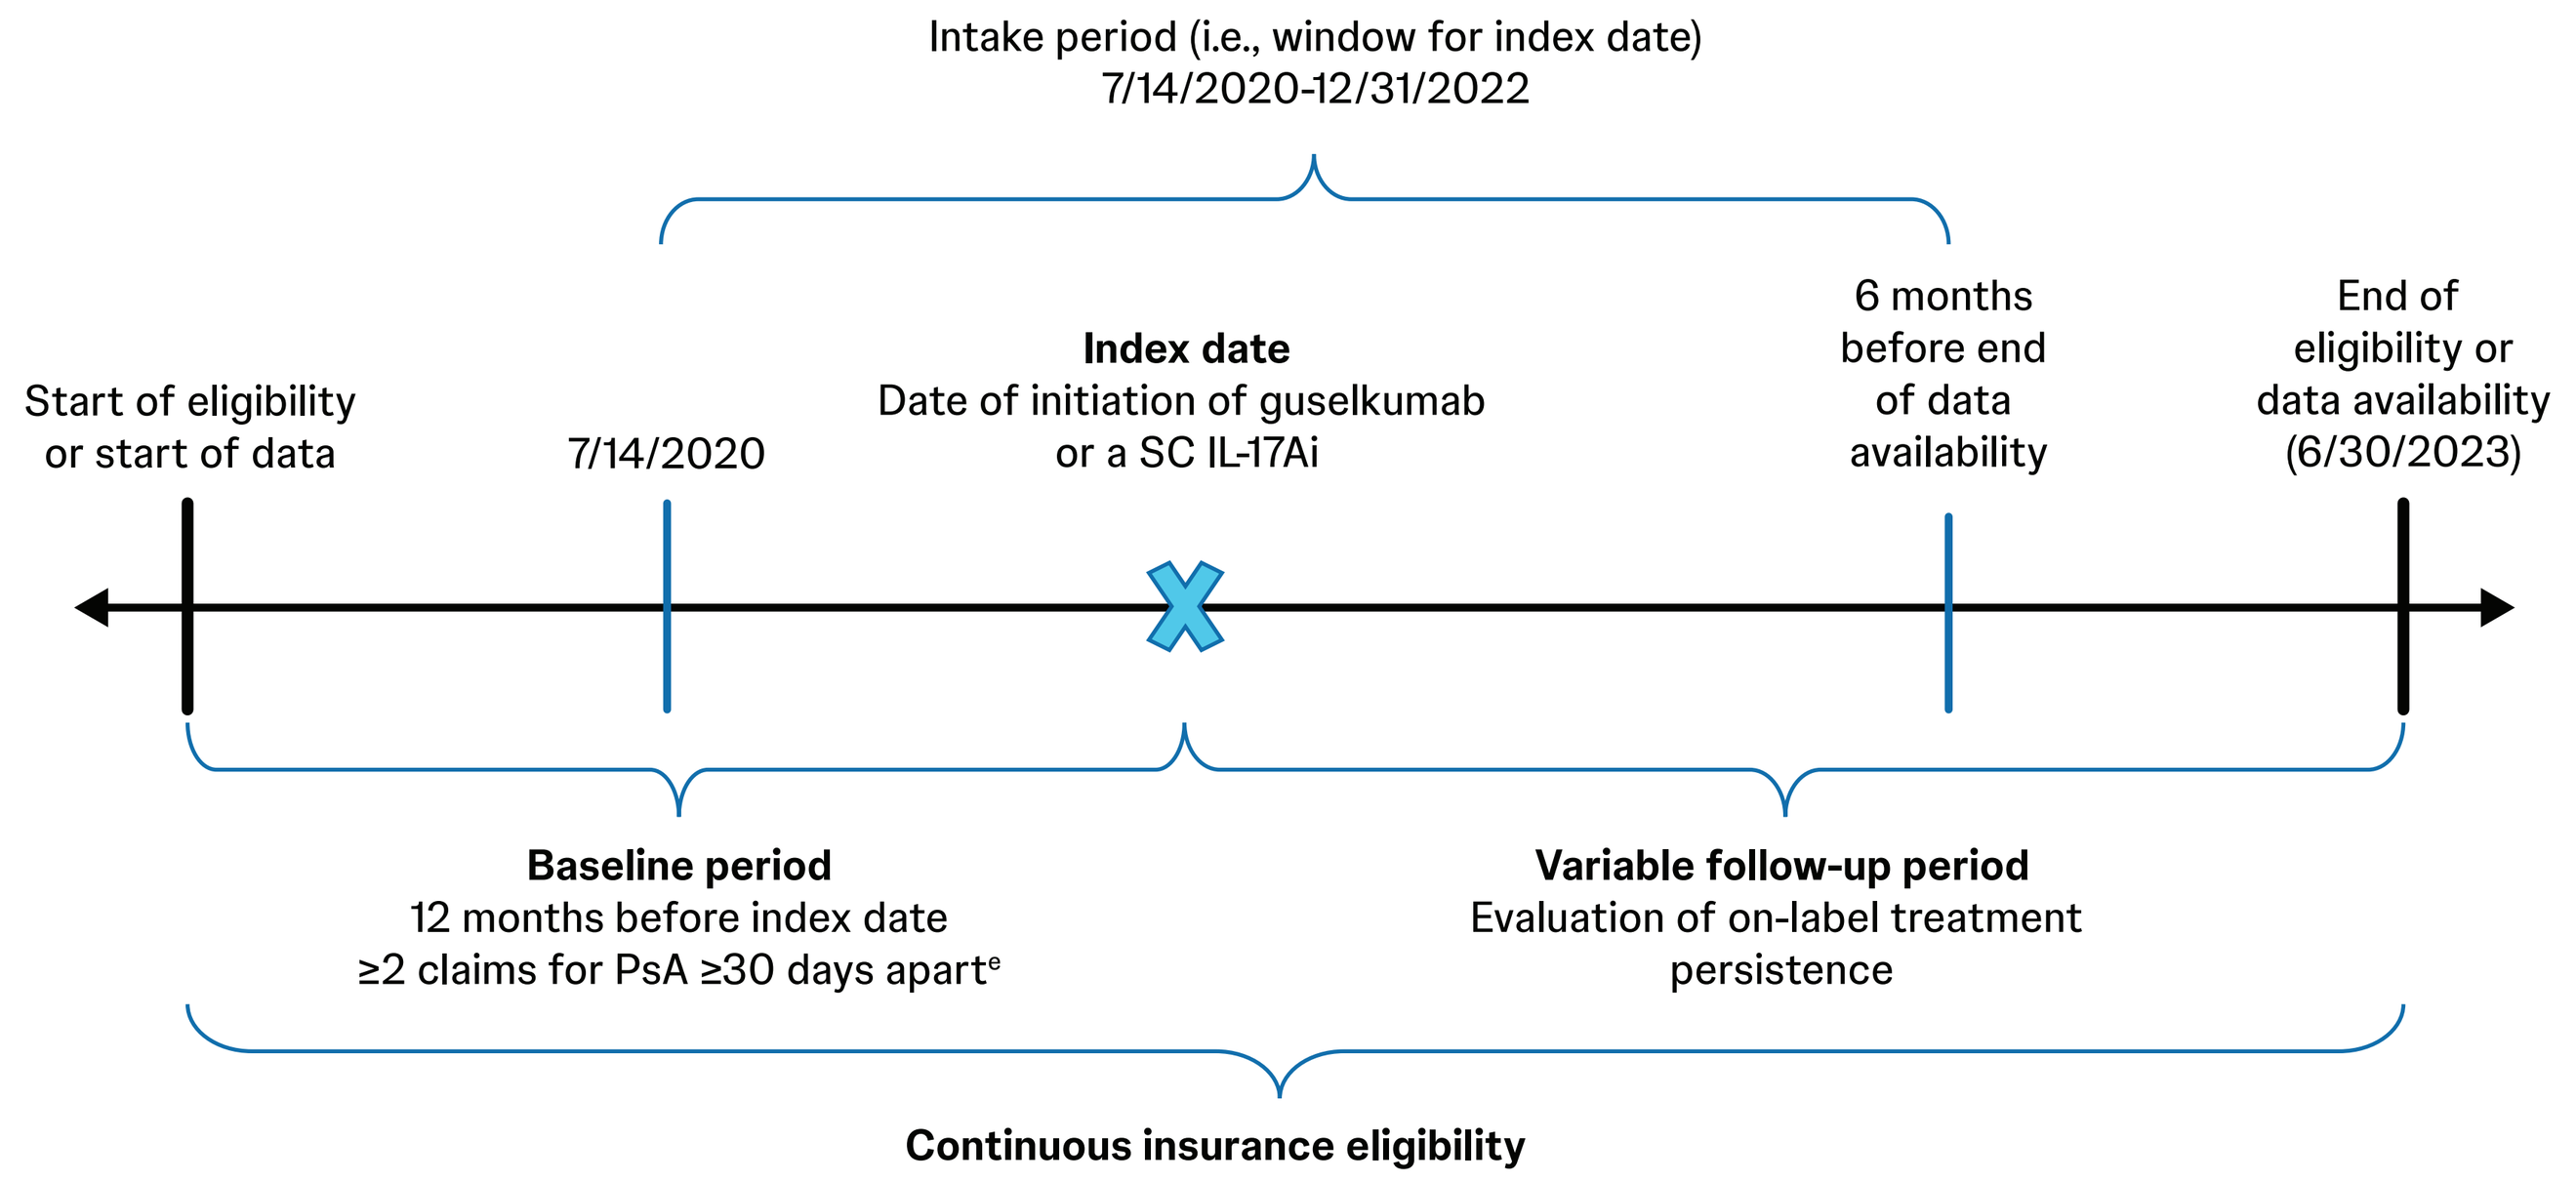
Intake period (i.e., window for index date)
7/14/2020-12/31/2022
Start of eligibility or start of data
7/14/2020
Index date
Date of initiation of guselkumab or a SC IL-17Ai
6 months before end of data availability
End of eligibility or data availability (6/30/2023)
Baseline period
12 months before index date
≥2 claims for PsA ≥30 days apart[e]
Variable follow-up period
Evaluation of on-label treatment persistence
Continuous insurance eligibility

[a]The IQVIA™ Health Plan Claims Data is comprised of fully adjudicated claims for inpatient and outpatient services, and outpatient prescription drugs, offering a diverse representation of geographic zones, employers, payers, providers, and therapy areas. [b]A validated algorithm for identifying patients with PsA in US claims data was used: ≥2 claims with a PsA diagnosis (ICD-10-CM: L40.5x) ≥30 days apart and ≥1 prescription claim for a PsA-related medications (i.e., guselkumab or SC IL-17Ai). [c]Patients could be bio-naïve or bio-experienced during baseline but were naïve to treatment with guselkumab or SC IL-17Ai agents. [d]Patients in the SC IL-17Ai cohort were newly initiated within the class. [e]Diagnoses for PsA include claims on the index date.

Patient Selection

- Index date: 1st GUS or SC IL-17Ai claim during intake period (7/14/2020-12/31/2022)[a]
- PsA pt identification: ≥2 PsA Dx (ICD-10-CM code L40.5x) ≥30 days apart within 12 months prior to or on the index date, and ≥1 claim for either GUS or first SC IL-17Ai[3]
- ≥12 months of continuous health insurance eligibility before index date
- ≥18 years of age
- No claims for other conditions for which GUS or IL-17Ai are approved or other potentially confounding diseases[b]

Dx=Diagnoses; ICD-10-CM=International Classification of Disease, 10th revision, Clinical Modification. [a]Pts could not have claims for >1 index agent on index date. [b]Pts were excluded if they had a claim for ankylosing spondylitis, other inflammatory arthritides, other spondylopathies, rheumatoid arthritis, systemic connective tissue disorders, relapsing polychondritis, unclassified connective tissue disease, hidradenitis suppurativa, inflammatory bowel disease, or uveitis in the 12-month baseline period preceding the index date.

Censoring and Imputations

Censoring: On earliest of first off-label claim or last day of index agent supply preceding end of follow-up period if discontinuation was not observed

Days of supply imputation rule	GUS	SC IL-17Ai
Medical Claims[1,4,5]		
1st claim	28 days	N/A[b]
2nd + claims	56 days	N/A[b]
Pharmacy Claims		
1st claim	28 days	No imputation[c]
2nd + claims	Based on time to next claim[a]	No imputation[c]

[a]28 days if time to next claim <42 days; 56 days if time to next claim 42-70 days; 84 days if time to next claim >70 days; if there is no next claim, days of supply of the previous claim was carried forward or imputed as 56 days if the original value was missing or if this was the second claim; no imputation for claims with days supply 56-60 or >60. [b]There is no Healthcare Common Procedure Coding System code for SC IL-17Ai in medical claims. [c]Pharmacy claims for SC IL-17Ai are typically consistent with approved labeling; therefore, reported days supply was used for SC IL-17Ai and no imputation was performed.

Statistical Analyses

- **Baseline demographic and disease characteristics (12 months pre-index):**
 - Balanced between the GUS and SC IL-17Ai cohorts using propensity score-weighting (overlap weights)
- **On-label persistence up to 24 months post-index:**
 - No treatment discontinuation or dose modification relative to US FDA-approved labeling
 - Proportion of pts determined using ***weighted*** KM curves
 - GUS vs SC IL-17Ai cohorts compared using ***weighted*** Cox proportional hazard models

Days between administrations[a]	GUS	SC IL-17Ai
Primary analysis		
2x[1,4,5]	112 days	56 days
Sensitivity analyses		
1x[1,4,5]	56 days	28 days
Fixed gap	112 days	112 days

KM=Kaplan-Meier. [a]Primary analysis was conducted based on 2x the FDA maintenance interval between administration per label after induction. Sensitivity analyses were conducted based on 1x the FDA maintenance interval between administration per label after induction as well as a fixed discontinuation gap of 112 days.

Key Takeaways

- **First real-world claims data analysis of treatment persistence over 24 months between active PsA pts newly initiated on GUS vs initial SC IL-17Ai per US FDA-approved labeling**
- **Pts in the GUS vs SC IL-17Ai cohort were significantly (~1.5x) more likely to remain persistent on treatment through 24 months**
- **Higher long-term on-label persistence may improve disease management outcomes, including functional status and quality of life, in pts with active PsA initiating GUS[6]**

Results

The GUS and SC IL-17Ai cohorts included 849 and 2,601 pts, respectively

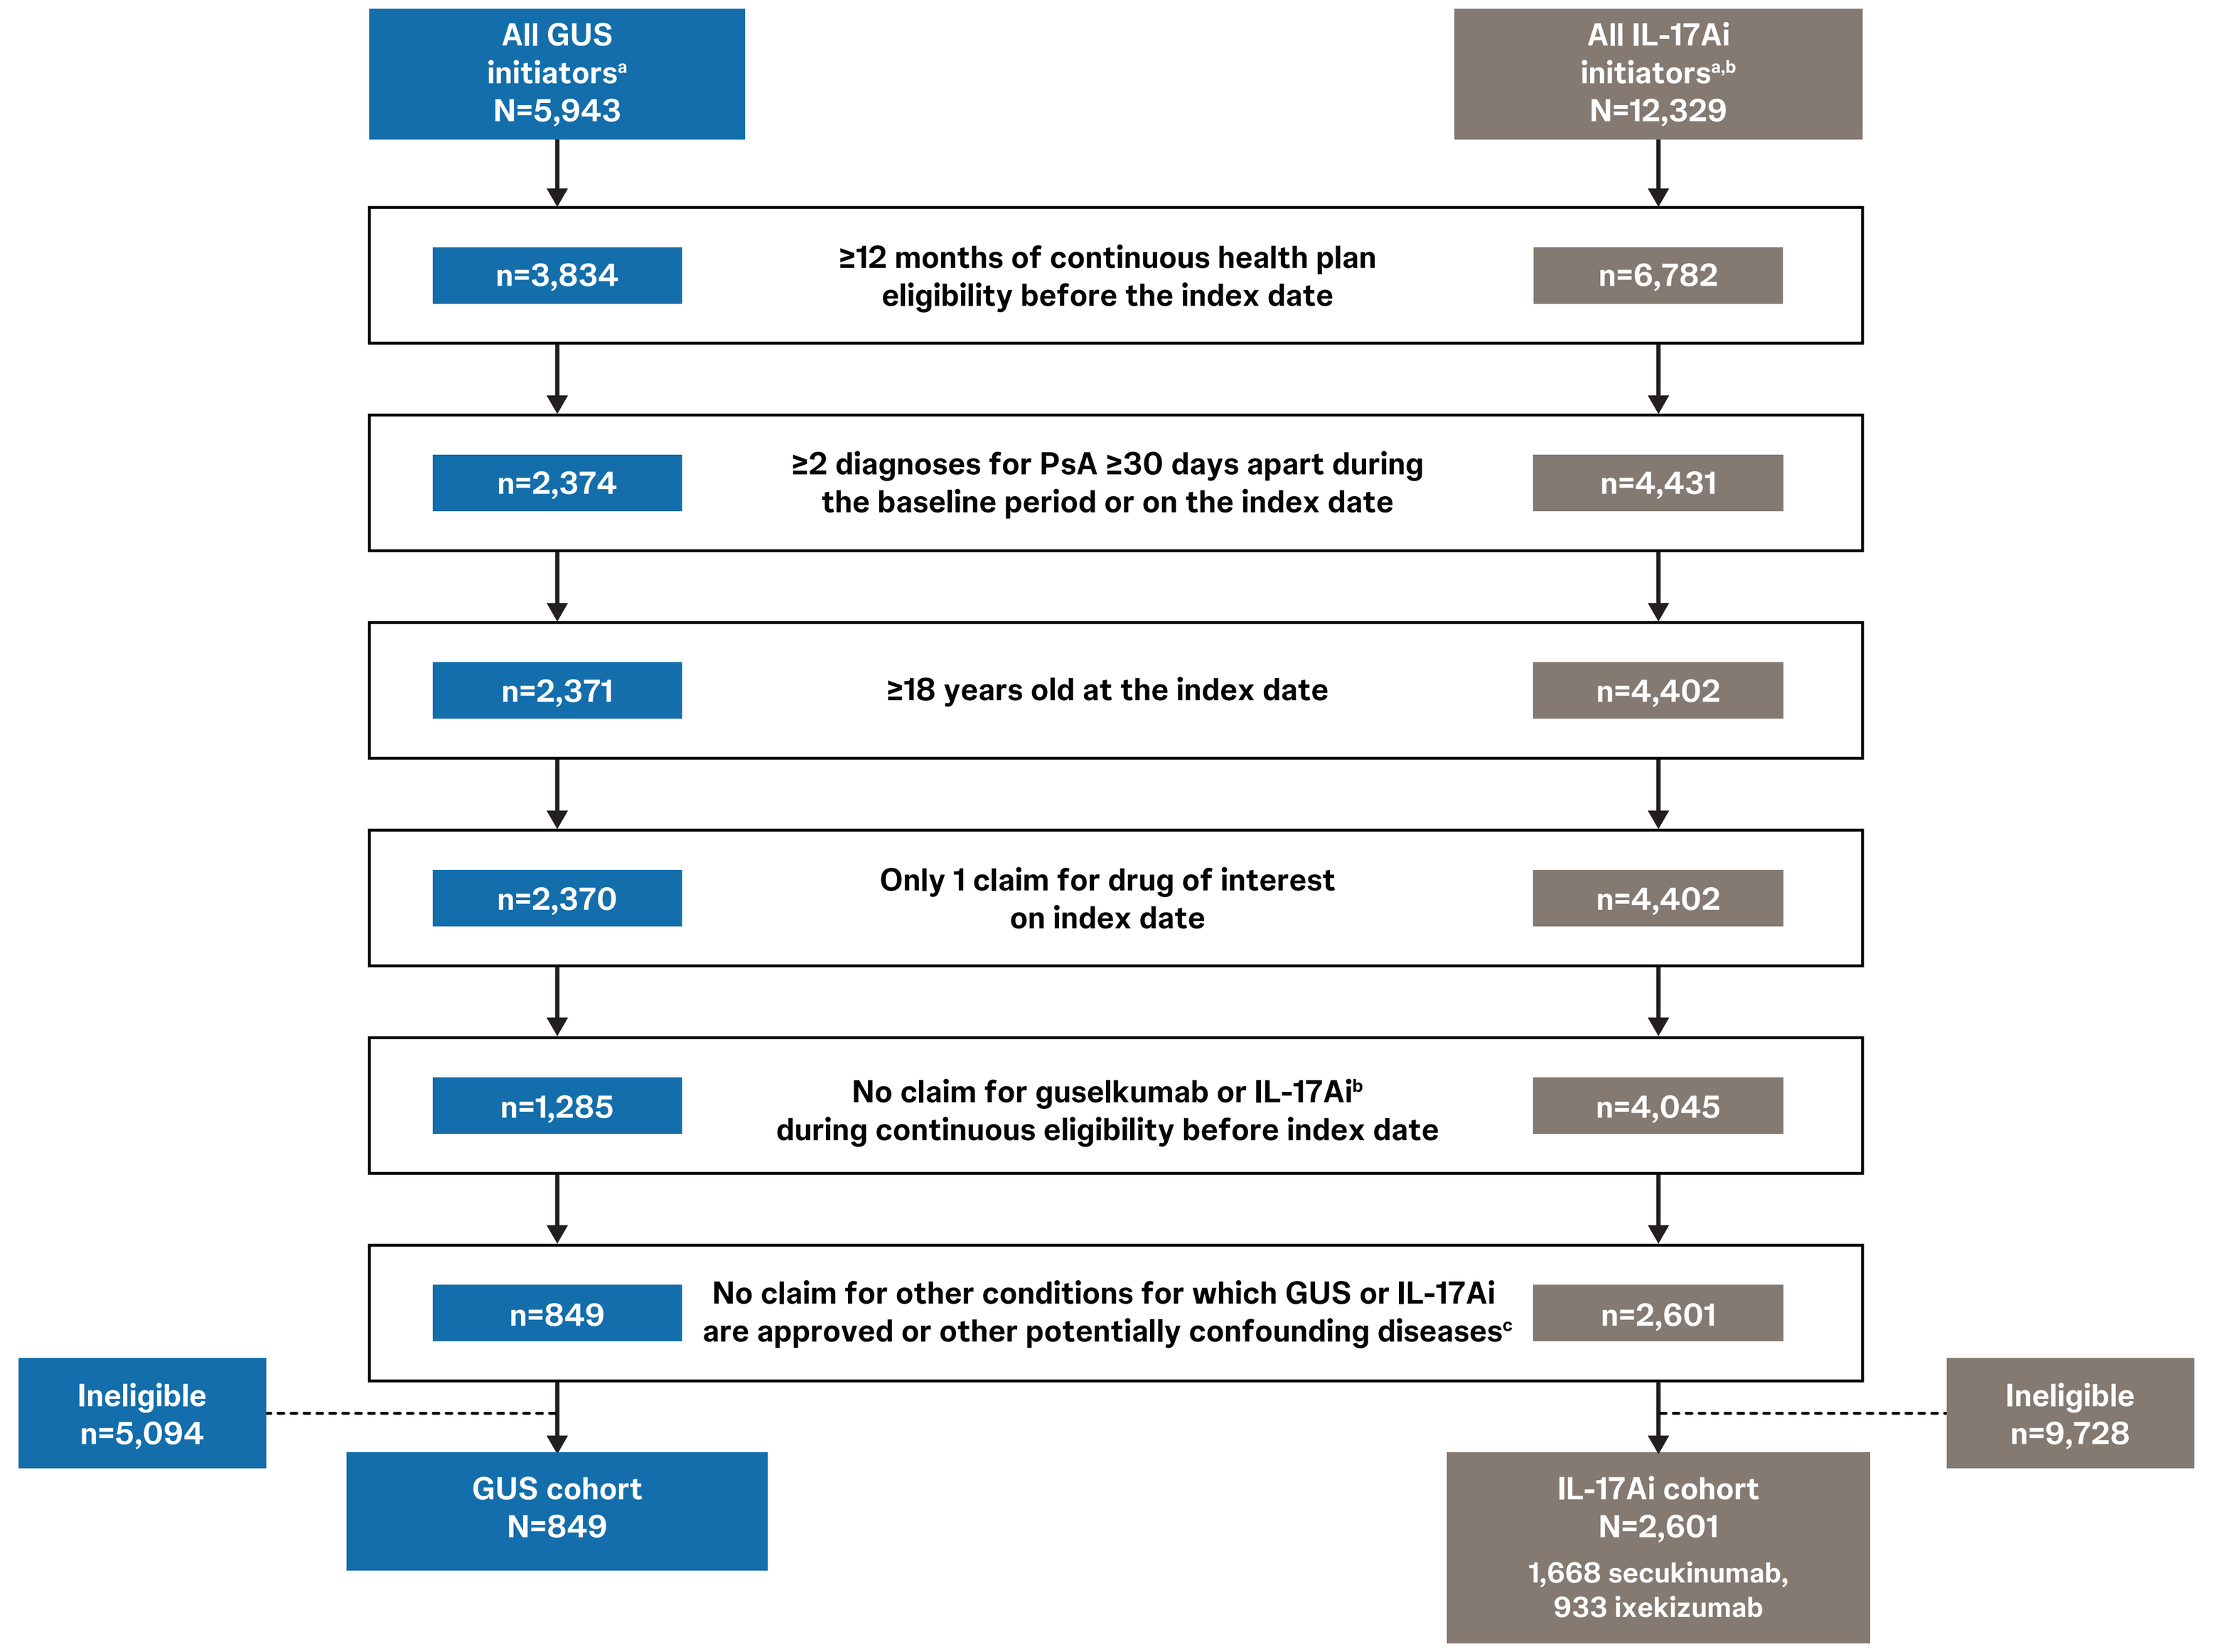
All GUS initiators[a]
N=5,943
All IL-17Ai initiators[a,b]
N=12,329
n=3,834
≥12 months of continuous health plan eligibility before the index date
n=6,782
n=2,374
≥2 diagnoses for PsA ≥30 days apart during the baseline period or on the index date
n=4,431
n=2,371
≥18 years old at the index date
n=4,402
n=2,370
Only 1 claim for drug of interest on index date
n=4,402
n=1,285
No claim for guselkumab or IL-17Ai[b] during continuous eligibility before index date
n=4,045
n=849
No claim for other conditions for which GUS or IL-17Ai are approved or other potentially confounding diseases[c]
n=2,601
Ineligible
n=5,094
Ineligible
n=9,728
GUS cohort
N=849
IL-17Ai cohort
N=2,601
1,668 secukinumab, 933 ixekizumab

[a]1st GUS or SC IL-17Ai claim during intake period (7/14/2020-12/31/2022). [b]The SC IL-17Ai cohort is defined as pts with an index claim for an SC IL-17Ai (ie, ixekinumab or secukinumab). [c]Assessed during the 12-month baseline period.

Weighted baseline demographic and clinical characteristics were similar between the GUS and SC IL-17Ai cohorts

- 57.4.% in the GUS cohort and 67.5% in the SC IL-17Ai cohort had received ≥1 bDMARD at any time before the index date[a]

Table 1. Weighted Baseline Demographics and Clinical Characteristics[b]

	GUS (N=849)	SC IL-17Ai (N=2,601)
Demographics		
Age at index date (years), Mean ± SD [median]	49.7 ± 11.0 [50.9]	49.6 ± 11.3 [50.8]
Female	59.4	59.4
Insurance type at index date		
Preferred provider organization	78.0	78.5
Health maintenance organization	11.0	11.0
Other[c]	11.0	10.5
Year of index date		
2020	11.6	11.6
2021	39.7	39.7
2022	48.7	48.7
Characteristics		
Months between latest observed PsA diagnosis and index date, Mean ± SD [median]	1.3 ± 1.6 [0.7]	1.3 ± 1.4 [0.8]
Quan-CCI, Mean ± SD [median]	0.6 ± 1.3 [0.0]	0.6 ± 1.3 [0.0]
Comorbidities		
Hyperlipidemia	34.8	36.6
Osteoarthritis	28.7	31.3
Diabetes	14.3	15.0
Peripheral vascular disease	2.7	2.2
Psoriasis	84.5	84.5
Smoking	9.9	11.5
Medication Use[d], %		
bDMARDs[e]	50.5	50.5
0	49.5	49.5
1	44.0	43.7
≥2	6.6	6.8
csDMARDs[f]	25.7	27.0
tsDMARDs[g]	21.9	21.9
Corticosteroids	72.5	71.5

Data are % unless otherwise noted. bDMARD=biologic disease-modifying antirheumatic drug; csDMARD=conventional synthetic DMARD; Quan-CCI=Quan Charlson Comorbidity Index; SD=standard deviation; tsDMARD=targeted synthetic DMARD. [a]Unweighted values. [b]Propensity score using overlap weighting. [c]Includes point-of-service, consumer directed health care, indemnity/traditional, and unknown plan type. [d]During 12 months before index date. [e]Includes IL-12/23i (ie, ustekinumab), anti-CTLA-4 (cytotoxic T lymphocyte-associated antigen, [ie, abatacept]), IL-23i (ie, risankizumab), SC TNFi (ie, adalimumab, certolizumab pegol, etanercept, golimumab), and IV TNFi (ie, infliximab, infliximab biosimilars, and IV golimumab). [f]Includes methotrexate, leflunomide, cyclosporine, mycophenolate, and azathioprine. [g]Includes apremilast, deucravacitinib, and Janus kinase inhibitors (ie, upadacitinib, baricitinib, and tofacitinib).

Pts in the GUS vs SC IL-17Ai cohort were significantly (1.5x) more likely to remain persistent with on-label treatment through 24 months

- **% pts with on-label persistence at 24 months:** GUS (44.9%) vs SC IL-17Ai (35.0%)
- **Median time to discontinuation:** GUS (20.9 months) vs SC IL-17Ai (12.2 months)
- **Sensitivity analyses:**
 - 1x FDA maintenance gap: HR (95% CI) =1.54 (1.36; 1.75); p<0.001
 - Fixed gap (112 days): HR (95% CI) =1.09 (0.94; 1.27); p=0.252

Primary KM Analysis (2x duration) of On-Label Persistence in Weighted GUS and SC IL-17Ai Cohorts[a,b]

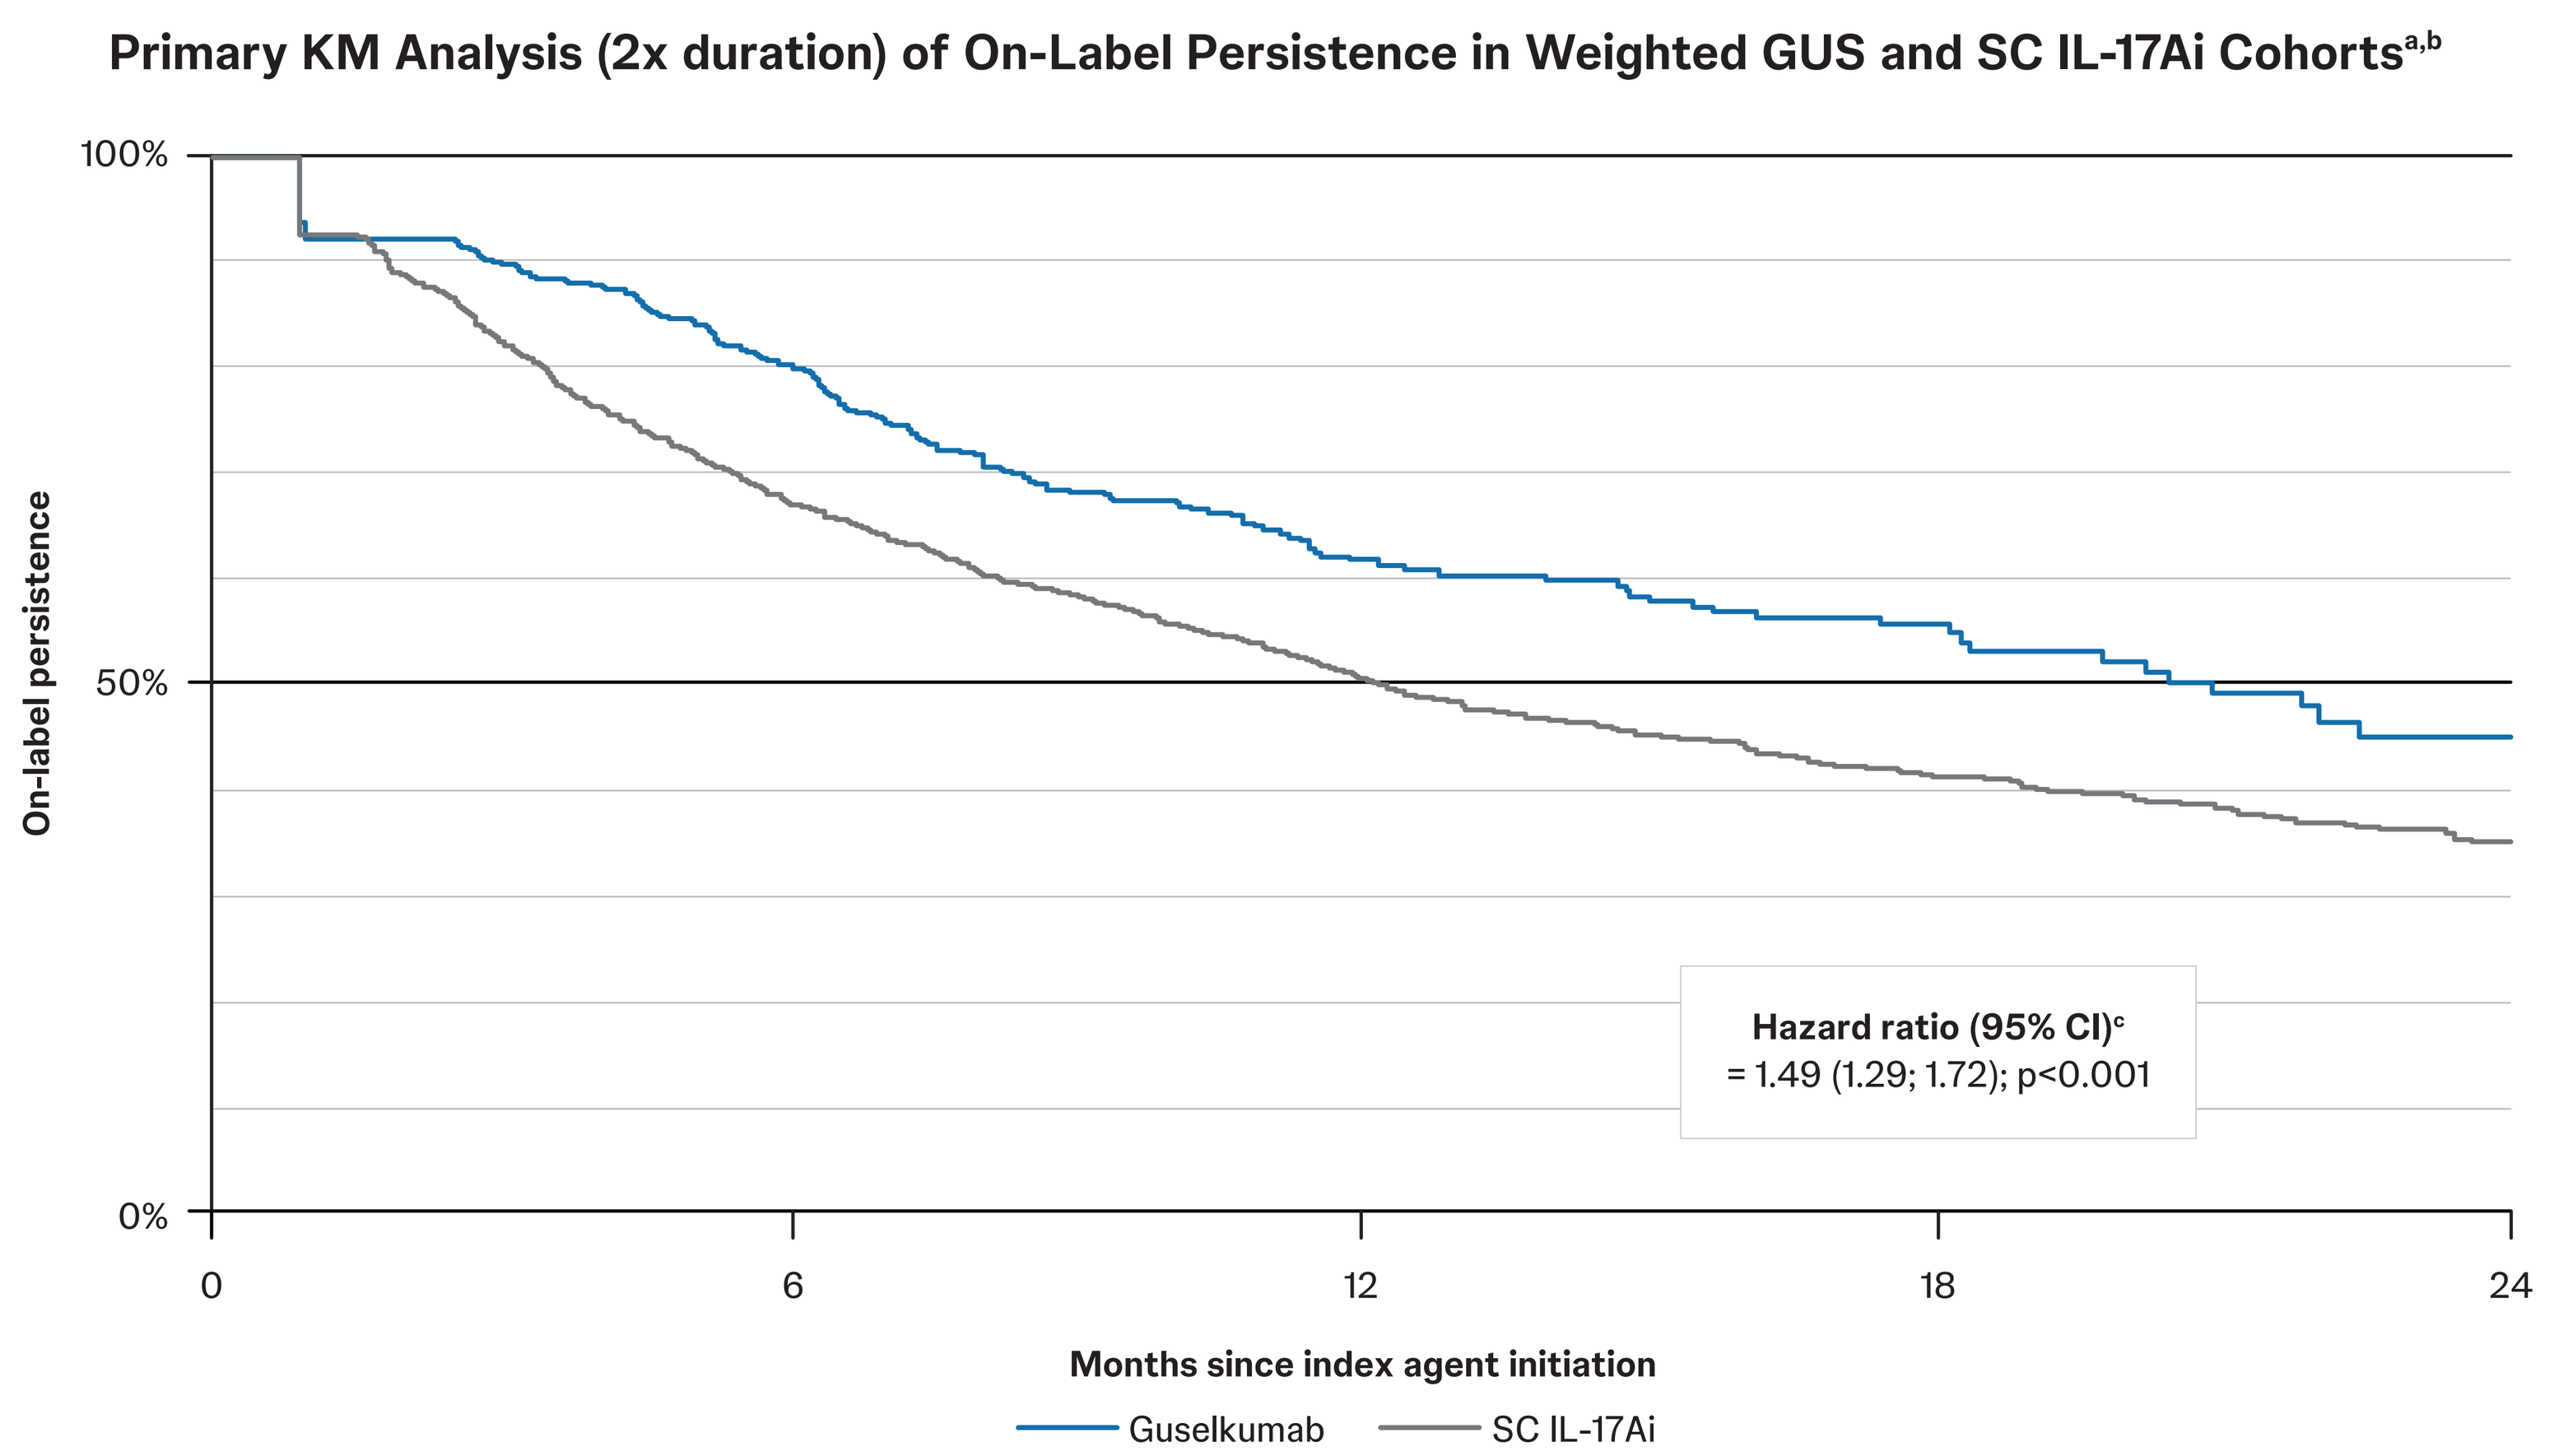
100%
50%
0%
On-label persistence
0
6
12
18
24
Months since index agent initiation
Guselkumab
SC IL-17Ai
Hazard ratio (95% CI)[c]
= 1.49 (1.29; 1.72); p<0.001

CI=confidence interval; [a]Discontinuation was defined as having a gap in treatment of more than twice the duration of days of supply for a claim (i.e., 2 x 56 = 112 days for guselkumab or 2 x 28 = 56 days for IL-17Ai). [b]Patients with dose changes inconsistent with FDA-approved dosing were censored as of the first dose change. [c]A weighted Cox proportional hazards model was used to compare on-label persistence between cohorts.

GUS was associated with significantly higher on-label persistence vs SC IL-17Ai at each time point assessed (6/12/18/24 months)

Table 2. On-label persistence through 24 months in weighted GUS and SC IL-17Ai cohorts[a]

Primary analysis (2x duration)

Cox proportional hazards model[b]	6 months	12 months	18 months	24 months
Pts at risk, n (%)[c]				
GUS (N=849)	440 (51.8)	179 (21.1)	80 (9.5)	26 (3.1)
SC IL-17Ai (N=2,601)	980 (37.7)	460 (17.7)	225 (8.6)	106 (4.1)
Hazard ratios (95% CI)	1.75 (1.45; 2.12)	1.50 (1.29; 1.75)	1.53 (1.32; 1.77)	1.49 (1.29; 1.72)
Chi-square p-value	<0.001	<0.001	<0.001	<0.001
KM Persistence, % (95% CI)				
GUS	80.3 (74.8; 84.8)	61.9 (55.4; 67.7)	55.7 (47.8; 62.9)	44.9 (30.2; 58.6)
SC IL-17Ai	68.0 (64.3; 71.4)	50.5 (45.9; 55.0)	41.5 (35.7; 47.1)	35.0 (27.6; 42.6)
Log-rank test p-value	<0.001	<0.001	<0.001	<0.001

[a]Propensity score weights were used to obtain a balanced sample. Weights were estimated using a multivariable logistic regression model. Baseline covariates included several demographic and clinical characteristics. [b]Weighted Cox proportional hazard models were used to compare risk of discontinuation between the GUS and SC IL-17Ai cohorts. [c]Pts at risk of having the event are pts who have not had the event and have not been lost to follow-up at that point in time.

Strengths and Limitations

- Strengths:
 - A case-finding algorithm validated in US claims data was used to identify pts with PsA[3]
 - Baseline demographic and disease characteristics between the GUS and SC IL-17Ai cohorts were balanced using propensity score-weighting based on overlap weights, minimizing risk of potential confounding due to differences at baseline
 - The claims database assessed a large sample of commercially insured PsA pts in the US; results are likely to be highly generalizable to that population
- Limitations:
 - Results may not be generalizable to non-commercially insured US pts or pts outside of the US
 - Claims data do not ensure treatments are taken as prescribed
 - Claims data do not provide treatment effectiveness nor reasons for discontinuation
 - Days of supply in pharmacy claims data can be inaccurate due to coverage restrictions. Imputation is a valid approach that is often utilized in claims-based analyses, but may lead to misclassifications

PRESENTED BY: L Hou at CCR West; September 26th-29th, 2024; San Diego, CA, USA. **REFERENCES:** **1.** Tremfya: Package insert. Janssen Biotech, Inc.; 2022. **2.** Mease et al., *Adv Ther* 2024; submitted. **3.** Lee H. *Pharmacoepidemiol Drug Saf.* 2020;29:404-8. **4.** Cosentyx: Package insert. Novartis.; 2023. **5.** Taltz: Package insert. Eli Lilly.; 2022. **6.** Fitzgerald T. *Dermatol Ther.* 2023;13:1053-68. **ACKNOWLEDGMENTS:** Medical writing support was provided by Kristin L. Leppard, MS, of Janssen Scientific Affairs, LLC under the direction of the authors in accordance with Good Publication Practice guidelines (*Ann Intern Med*;2022 Sep;175(9):1298-1304). Sponsored by Janssen Research & Development, LLC, a Johnson & Johnson Company. **DISCLOSURES: PJM:** has received research grants from AbbVie, Acelyrin, Amgen, Bristol Myers Squibb, Eli Lilly, Janssen, Novartis, Pfizer, SUN, and UCB; consulting fees from AbbVie, Acelyrin, Aclaris, Amgen, Boehringer Ingelheim, Bristol Myers Squibb, Eli Lilly, Galapagos, Gilead, GlaxoSmithKline, Immagene, Janssen, Novartis, Pfizer, SUN, UCB, and Ventyx; speaker fees from AbbVie, Amgen, Eli Lilly, Janssen, Novartis, Pfizer, and UCB. **JW:** received research funding from AbbVie, Merck, and Pfizer; and consulting fees from AbbVie, Eli Lilly, Janssen, Novartis, and UCB. **TPF, SDC,** and **LH** are employees of Janssen Scientific Affairs, LLC, a Johnson & Johnson company and own stock in Johnson & Johnson. **BE, CR, SS, KY, YW, PL,** and **DP** are employees of Analysis Group, Inc., a consulting company that has received research funding from Janssen Scientific Affairs, LLC, a Johnson & Johnson company. **SS:** received research funding from Eli Lilly and consulting fees from AbbVie, Janssen, and UCB. **JFM:** is a consultant and/or investigator for AbbVie, Amgen, Astra-Zeneca, Biogen, Boehringer Ingelheim, Bristol-Myers Squibb, Dermavant, Eli Lilly, Incyte, Janssen, Leo Pharma, Moonlake, Novartis, Pfizer, Sanofi-Regeneron, Sun Pharma, and UCB.