P0950

Guselkumab Decreases Key Cellular Inflammatory Processes Across Ileum and Colon Tissue in Crohn's Disease

Dylan Richards,[1] Swati Venkat,[1] Darren Ruane,[1] Martha Zeeman,[1] Natalie A. Terry,[1] Marion Vetter,[1] Mario Gomez,[2] Daniel Cua,[1] Tom C. Freeman,[1] Bradford McRae,[1] Brian Feagan,[3] Walter Reinisch,[4] Patrick Branigan[1]

[1]Janssen Research & Development, LLC, Spring House, PA, USA; [2]Janssen Global Services, LLC, Horsham, PA, USA; [3]Alimentiv Inc., Western University, London, ON, Canada; [4]Division of Gastroenterology & Hepatology, Medical University of Vienna, Vienna, Austria.

Key Takeaways

- **IV induction treatment with GUS attenuated key proinflammatory and effector cytokines that are associated with the IL-23 pathway in CD**
- **Tissue transcriptomics demonstrated attenuation of key immune- and inflammatory-related processes across all tissue regions by GUS compared with PBO**
- **Segmental molecular changes reflect regional differences in endoscopic improvement**

Background

Crohn's disease (CD) is characterized by significant heterogeneity in phenotypic disease expression, making it challenging to define the molecular mechanisms associated with disease and response to therapy

In the GALAXI phase 2b induction study (NCT03466411), guselkumab (GUS), a dual-acting interleukin (IL)-23p19 subunit inhibitor, induced greater clinical and endoscopic improvement versus placebo (PBO) at Week 12, with a safety profile consistent with known indications[1]

Objective

Here, we provide a detailed evaluation of the cellular and molecular mode of action of GUS through Week 12 in participants with moderately to severely active CD from the GALAXI phase 2b induction study

Methods

Study Design

- GALAXI was a phase 2, double-blind, PBO- and active-controlled (ustekinumab served as a reference arm and was not included in this analysis), multicenter study of GUS in participants ≥18 years of age with moderately to severely active CD
- For the 12-week induction period, participants were randomized 1:1:1:1:1 to PBO (n = 61), GUS 200 mg intravenous (IV) every 4 weeks (Q4W; n = 61), GUS 600 mg IV Q4W (n = 63), GUS 1,200 mg IV Q4W (n = 61), or ustekinumab (~6 mg/kg IV followed by 90 mg subcutaneous at Week 8; n = 63)

Participant Samples

- Serum samples were evaluated at baseline (Week 0), Week 4, and Week 12 for proinflammatory and effector cytokines (serum amyloid A [SAA], C-reactive protein [CRP], IL-6, interferon gamma [IFNγ], IL-22, and IL-17A) from participants treated with GUS induction therapy (n = 131 combined) or PBO (n = 44) who had ≥1 paired sample at Week 0 with Week 4 or Week 12
- Baseline biopsies (n = 692) from 249 participants were obtained from the rectum, splenic flexure (SF), and terminal ileum. Transcriptional profiling was performed using 1 biopsy per segment (when available) with bulk RNA sequencing (RNAseq)
- A subset of participants from the combined GUS IV induction treatment and PBO groups (n = 217) with a history of stricture (investigator reported) or current ileal narrowing (Simple Endoscopic Score for CD [SES-CD] = 0-3) were evaluated for collagen formation (fragment of N-terminal type III collagen [PRO-C3]) and degradation (fragment of type III/IV collagen released by matrix metalloproteinase [C3M/C4M]) serum biomarkers

Molecular Activity Score

- An objective tissue-based molecular activity score (MAS) that is based on the association of tissue-based transcriptional modules with histology was used to identify inflamed biopsy samples[2]
 - Inflamed baseline tissue was used to assess segmental molecular disease profiles that were correlated with paired segmental histology scores (defined by Global Histologic Activity Score [GHAS]) and endoscopic severities (defined by SES-CD)
- Of the baseline biopsy samples evaluated, 313/692 (45.2%) were identified as inflamed

Inflamed Baseline Biopsy Sample Size/Total Biopsies (%)

	Baseline cohort (n = 692)
Ileum	114/222 (51.4%)
Colon (rectum + SF)	199/470 (42.3%)

Molecular Inflammation Represents the Majority of Involved Segments With Increased Specificity Compared With Endoscopy and Histology[2]

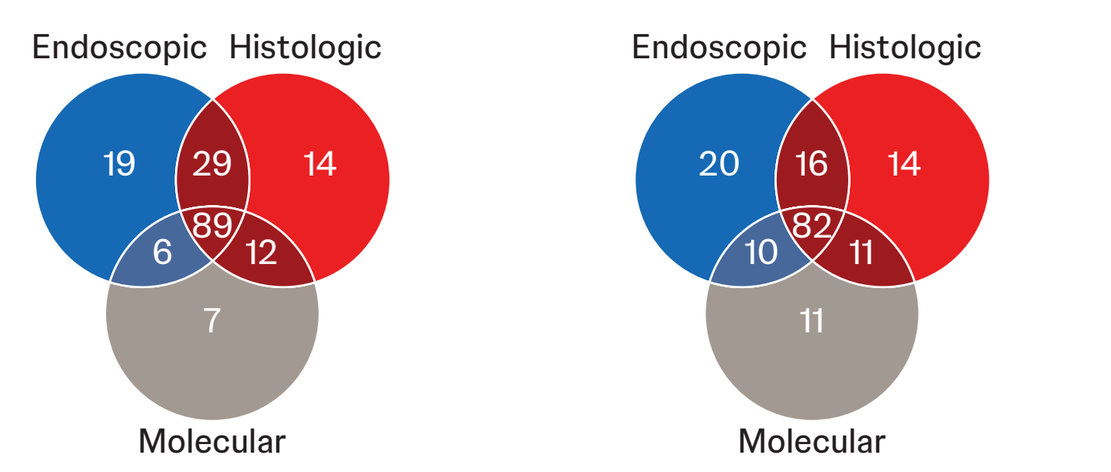
Ileum (n = 222)
Rectum (n = 241)
Endoscopic
Histologic
Molecular
19
29
14
89
6
12
7
20
16
14
82
10
11
11

Venn diagrams display the overlap of transcriptomic participant samples that are inflamed based on endoscopic (SES-CD >0), histologic (GHAS >0), and molecular scores (inflamed MAS).

- With segmental disease heterogeneity and biopsy sampling at pre-specified locations, biopsies are not always representative of disease extent, depth, and severity
- Local MAS per region showed the highest correlations with paired baseline GHAS values from adjacent biopsies (Spearman R: ileum, 0.70; SF, 0.63; rectum, 0.72), followed by SES-CD in each respective local segment (ileum, 0.58; rectum, 0.69)

GALAXI Biopsy Sampling Locations

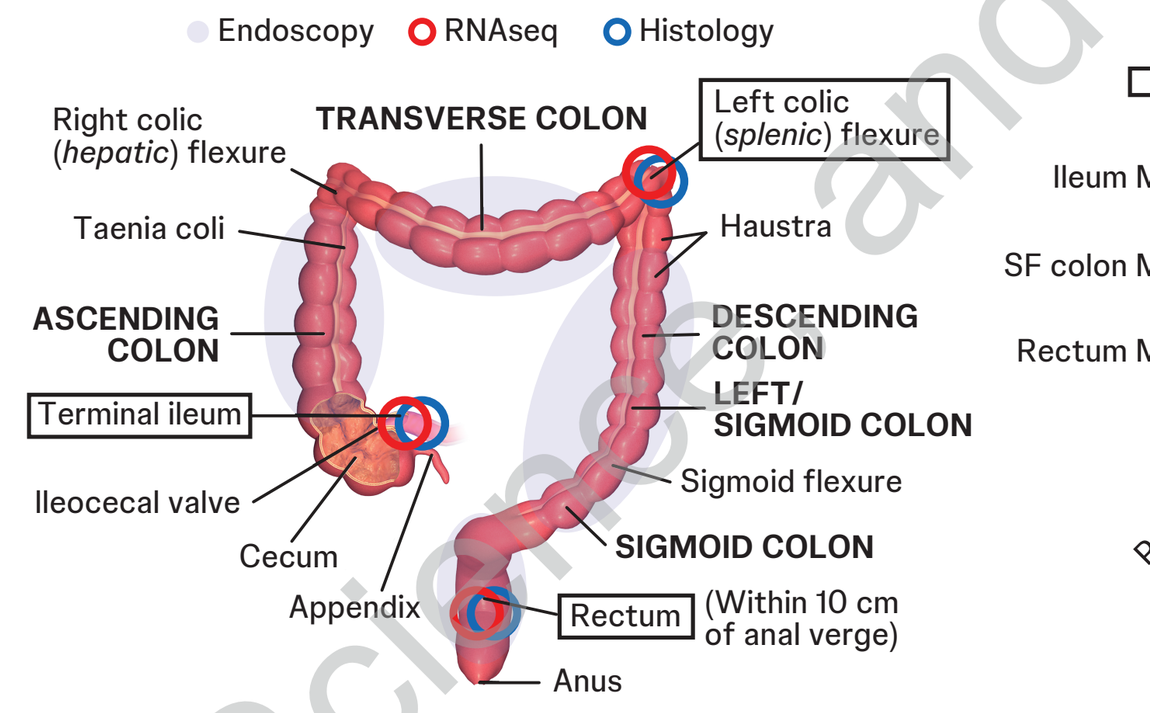
Endoscopy
RNAseq
Histology
Right colic (hepatic) flexure
TRANSVERSE COLON
Left colic (splenic) flexure
Taenia coli
Haustra
ASCENDING COLON
DESCENDING COLON
Terminal ileum
LEFT/ SIGMOID COLON
Ileocecal valve
Sigmoid flexure
Cecum
SIGMOID COLON
Appendix
Rectum
(Within 10 cm of anal verge)
Anus

*SF colon biopsy was taken from between the 2 regions (transverse and sigmoid colon).

Regional MAS Correlation With Regional Histologic and Endoscopic Scores

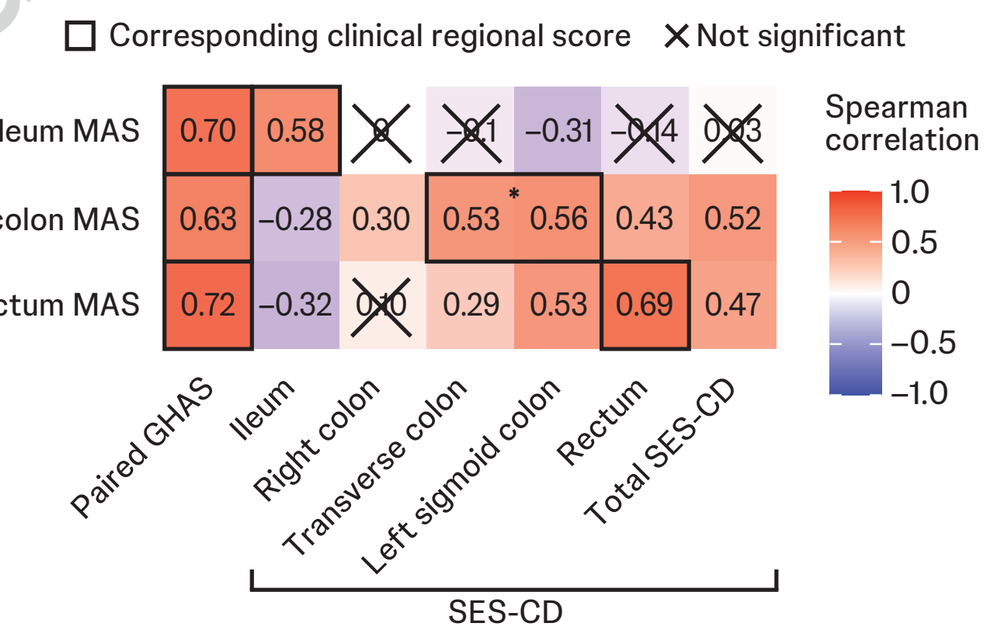
Corresponding clinical regional score
Not significant
Ileum MAS
SF colon MAS
Rectum MAS
0.70
0.58
−0.31
0.63
−0.28
0.30
0.53
0.56
0.43
0.52
0.72
−0.32
0.29
0.53
0.69
0.47
Paired GHAS
Ileum
Right colon
Transverse colon
Left sigmoid colon
Rectum
Total SES-CD
SES-CD
Spearman correlation
1.0
0.5
0
−0.5
−1.0

Results

GUS IV induction treatment reduced serum proinflammatory and effector cytokines associated with the IL-23 pathway as early as Week 4, which further declined through Week 12

Changes in Serum Inflammatory Proteins at Week 4 and Week 12 Compared With Baseline

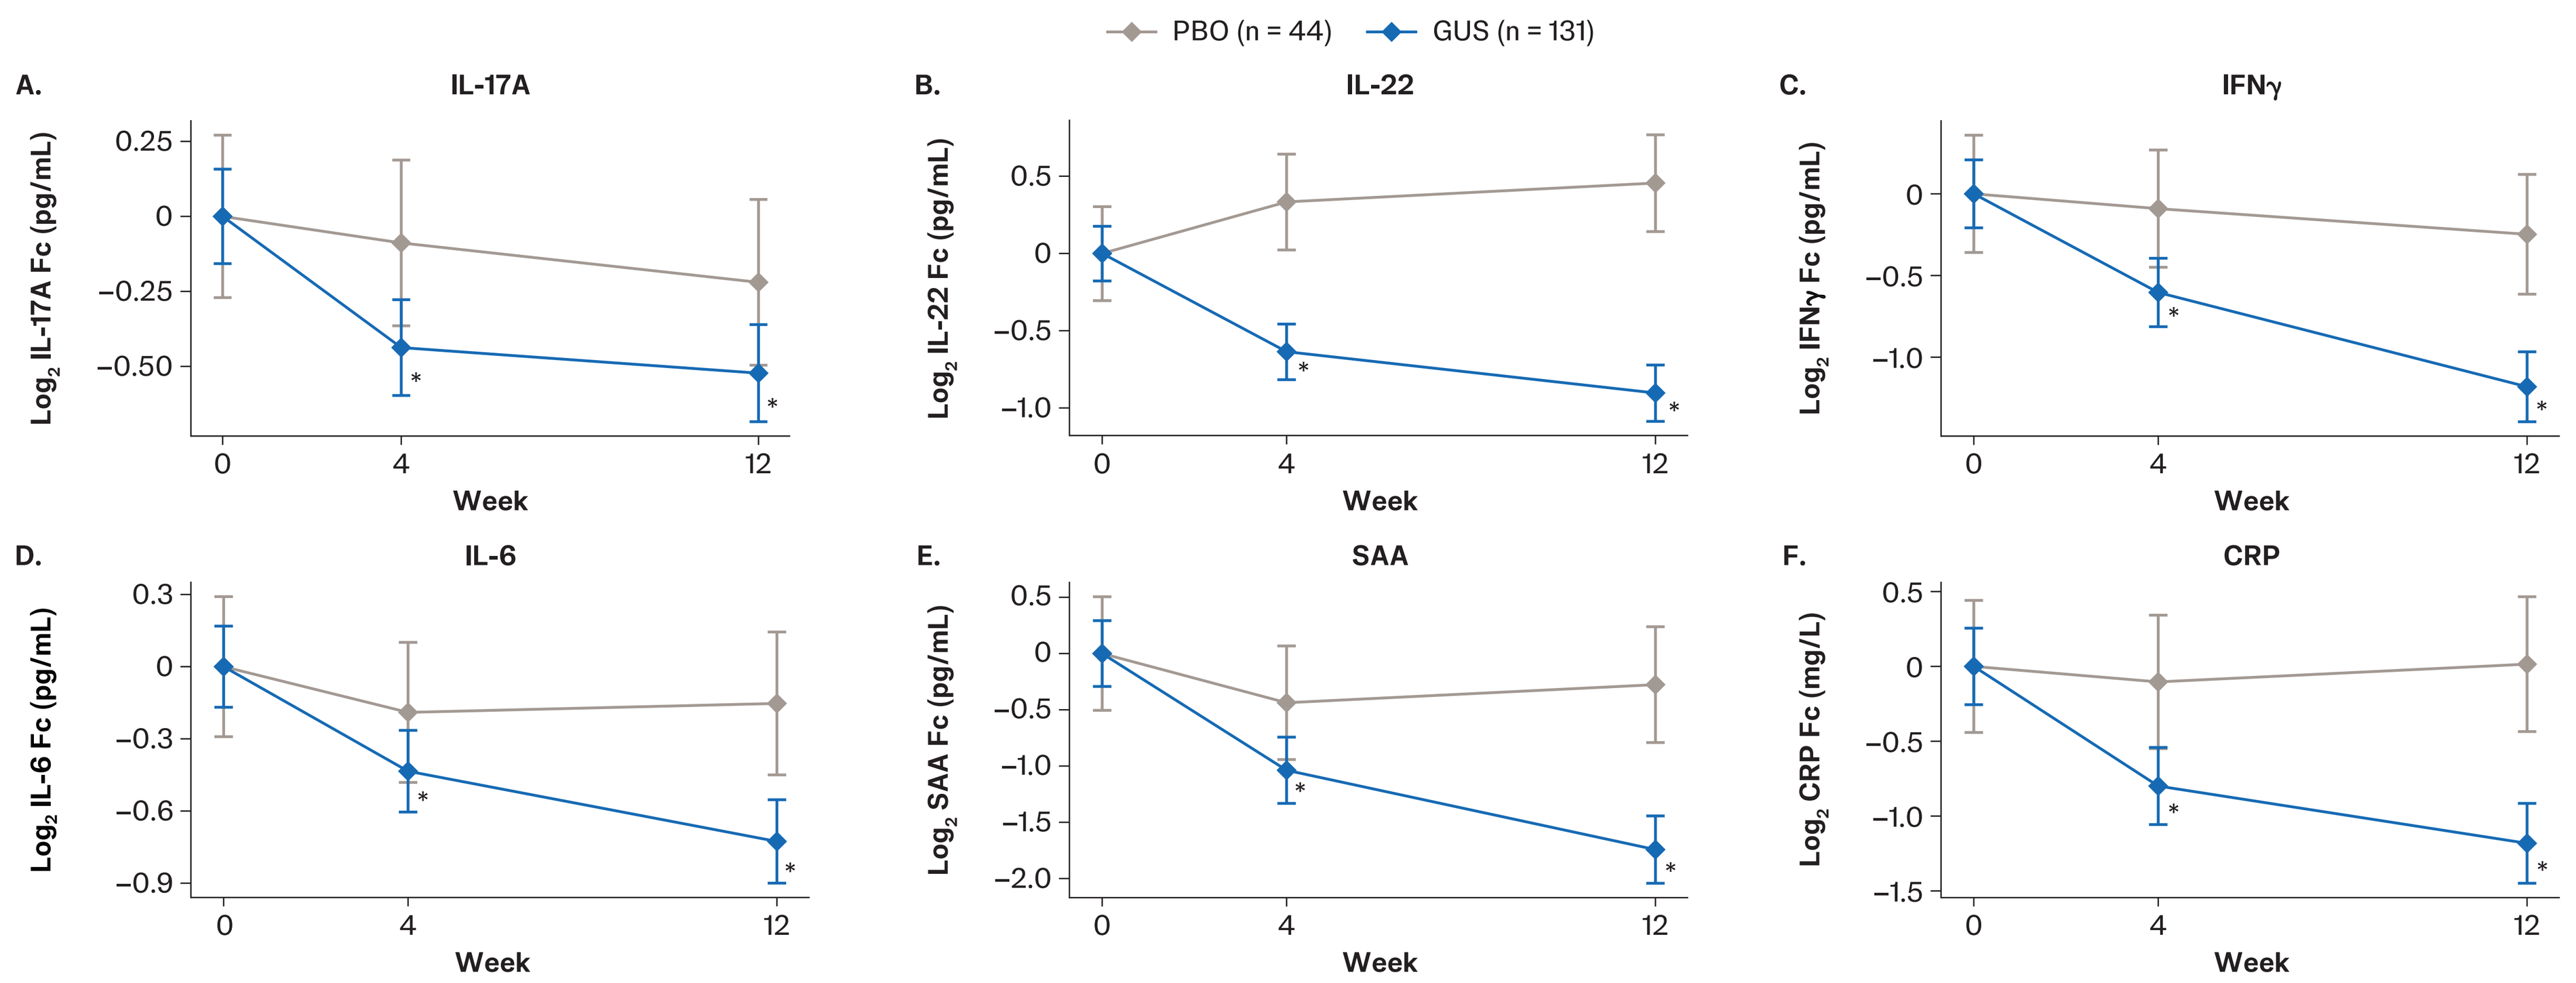
PBO (n = 44)
GUS (n = 131)
A. IL-17A
B. IL-22
C. IFNγ
D. IL-6
E. SAA
F. CRP
Log2 IL-17A Fc (pg/mL)
Log2 IL-22 Fc (pg/mL)
Log2 IFNγ Fc (pg/mL)
Log2 IL-6 Fc (pg/mL)
Log2 SAA Fc (pg/mL)
Log2 CRP Fc (mg/L)
Week
0
4
12

(A-B) IL-17A and IL-22 levels were assessed using a high-sensitivity Single Molecule Counting™ assay. (C-E) IFNγ, IL-6, and SAA were assessed using an electrochemiluminescence assay (Meso Scale Discovery). (F) CRP levels were assessed at a central laboratory (Labcorp). The y-axis represents the log_2 fold change in protein level.
*Nominal P <0.05 (treatment vs baseline contrasts per treatment, with participant as a random effect).

Compared with PBO at Week 12, GUS reduced tissue RNA-expression levels of IL17A, IL22, and IFNγ

- RNA expression was assessed in molecularly involved (MAS) biopsies from participants with endoscopic improvement (segmental SES-CD >0)
- Data represent ileal and colonic (rectum and SF) biopsies obtained from the combined GUS induction doses at baseline and Week 12 (n = 791) compared with PBO (n = 273)

Expression Profiles of the Genes Encoding IL-17A, IL-22, and IFNγ in CD Tissue as Determined by RNAseq

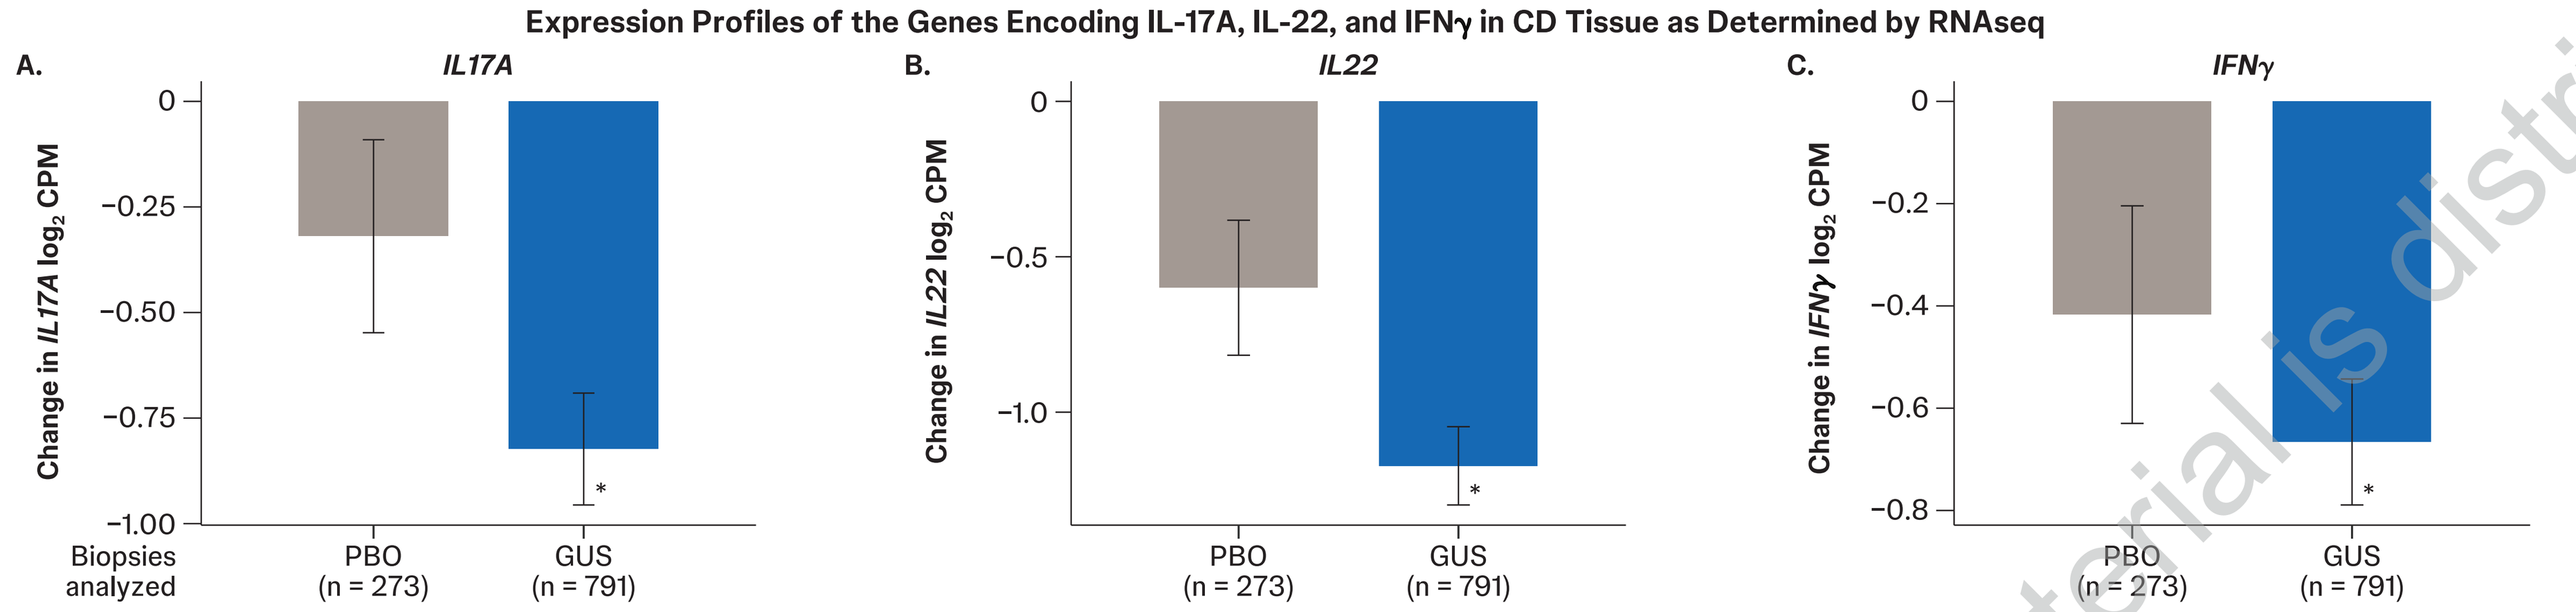
A. IL17A
B. IL22
C. IFNγ
Change in IL17A log2 CPM
Change in IL22 log2 CPM
Change in IFNγ log2 CPM
Biopsies analyzed
PBO (n = 273)
GUS (n = 791)

Paired biopsies were assessed from the combined GUS IV induction treatment (n = 140) and PBO (n = 51) groups. The y-axis represents the change in CPM mapped of RNAseq data for (A) IL17A, (B) IL22, and (C) IFNγ.
*P <0.05 (linear mixed model using time point treatment contrasts with segment location and drug exposure index as fixed effects and with participant as a random effect).
CPM, counts per million.

GUS significantly decreased cellular processes associated with the IL-23 pathway

- Transcriptional modules derived from a publicly available single-cell RNAseq dataset[3] were used to assess tissue-level gene-expression changes associated with specific cellular and immune processes at Week 12
 - The modules assessed represent the cellular processes associated with epithelial inflammation, inflammatory fibroblast and myeloid biology, interferon response, and the IL-23 pathway
- Segmental molecular change was greatest in the colon, followed by the rectum and ileum

Transcriptional Module Expression Profiles in CD Tissue as Determined by RNAseq

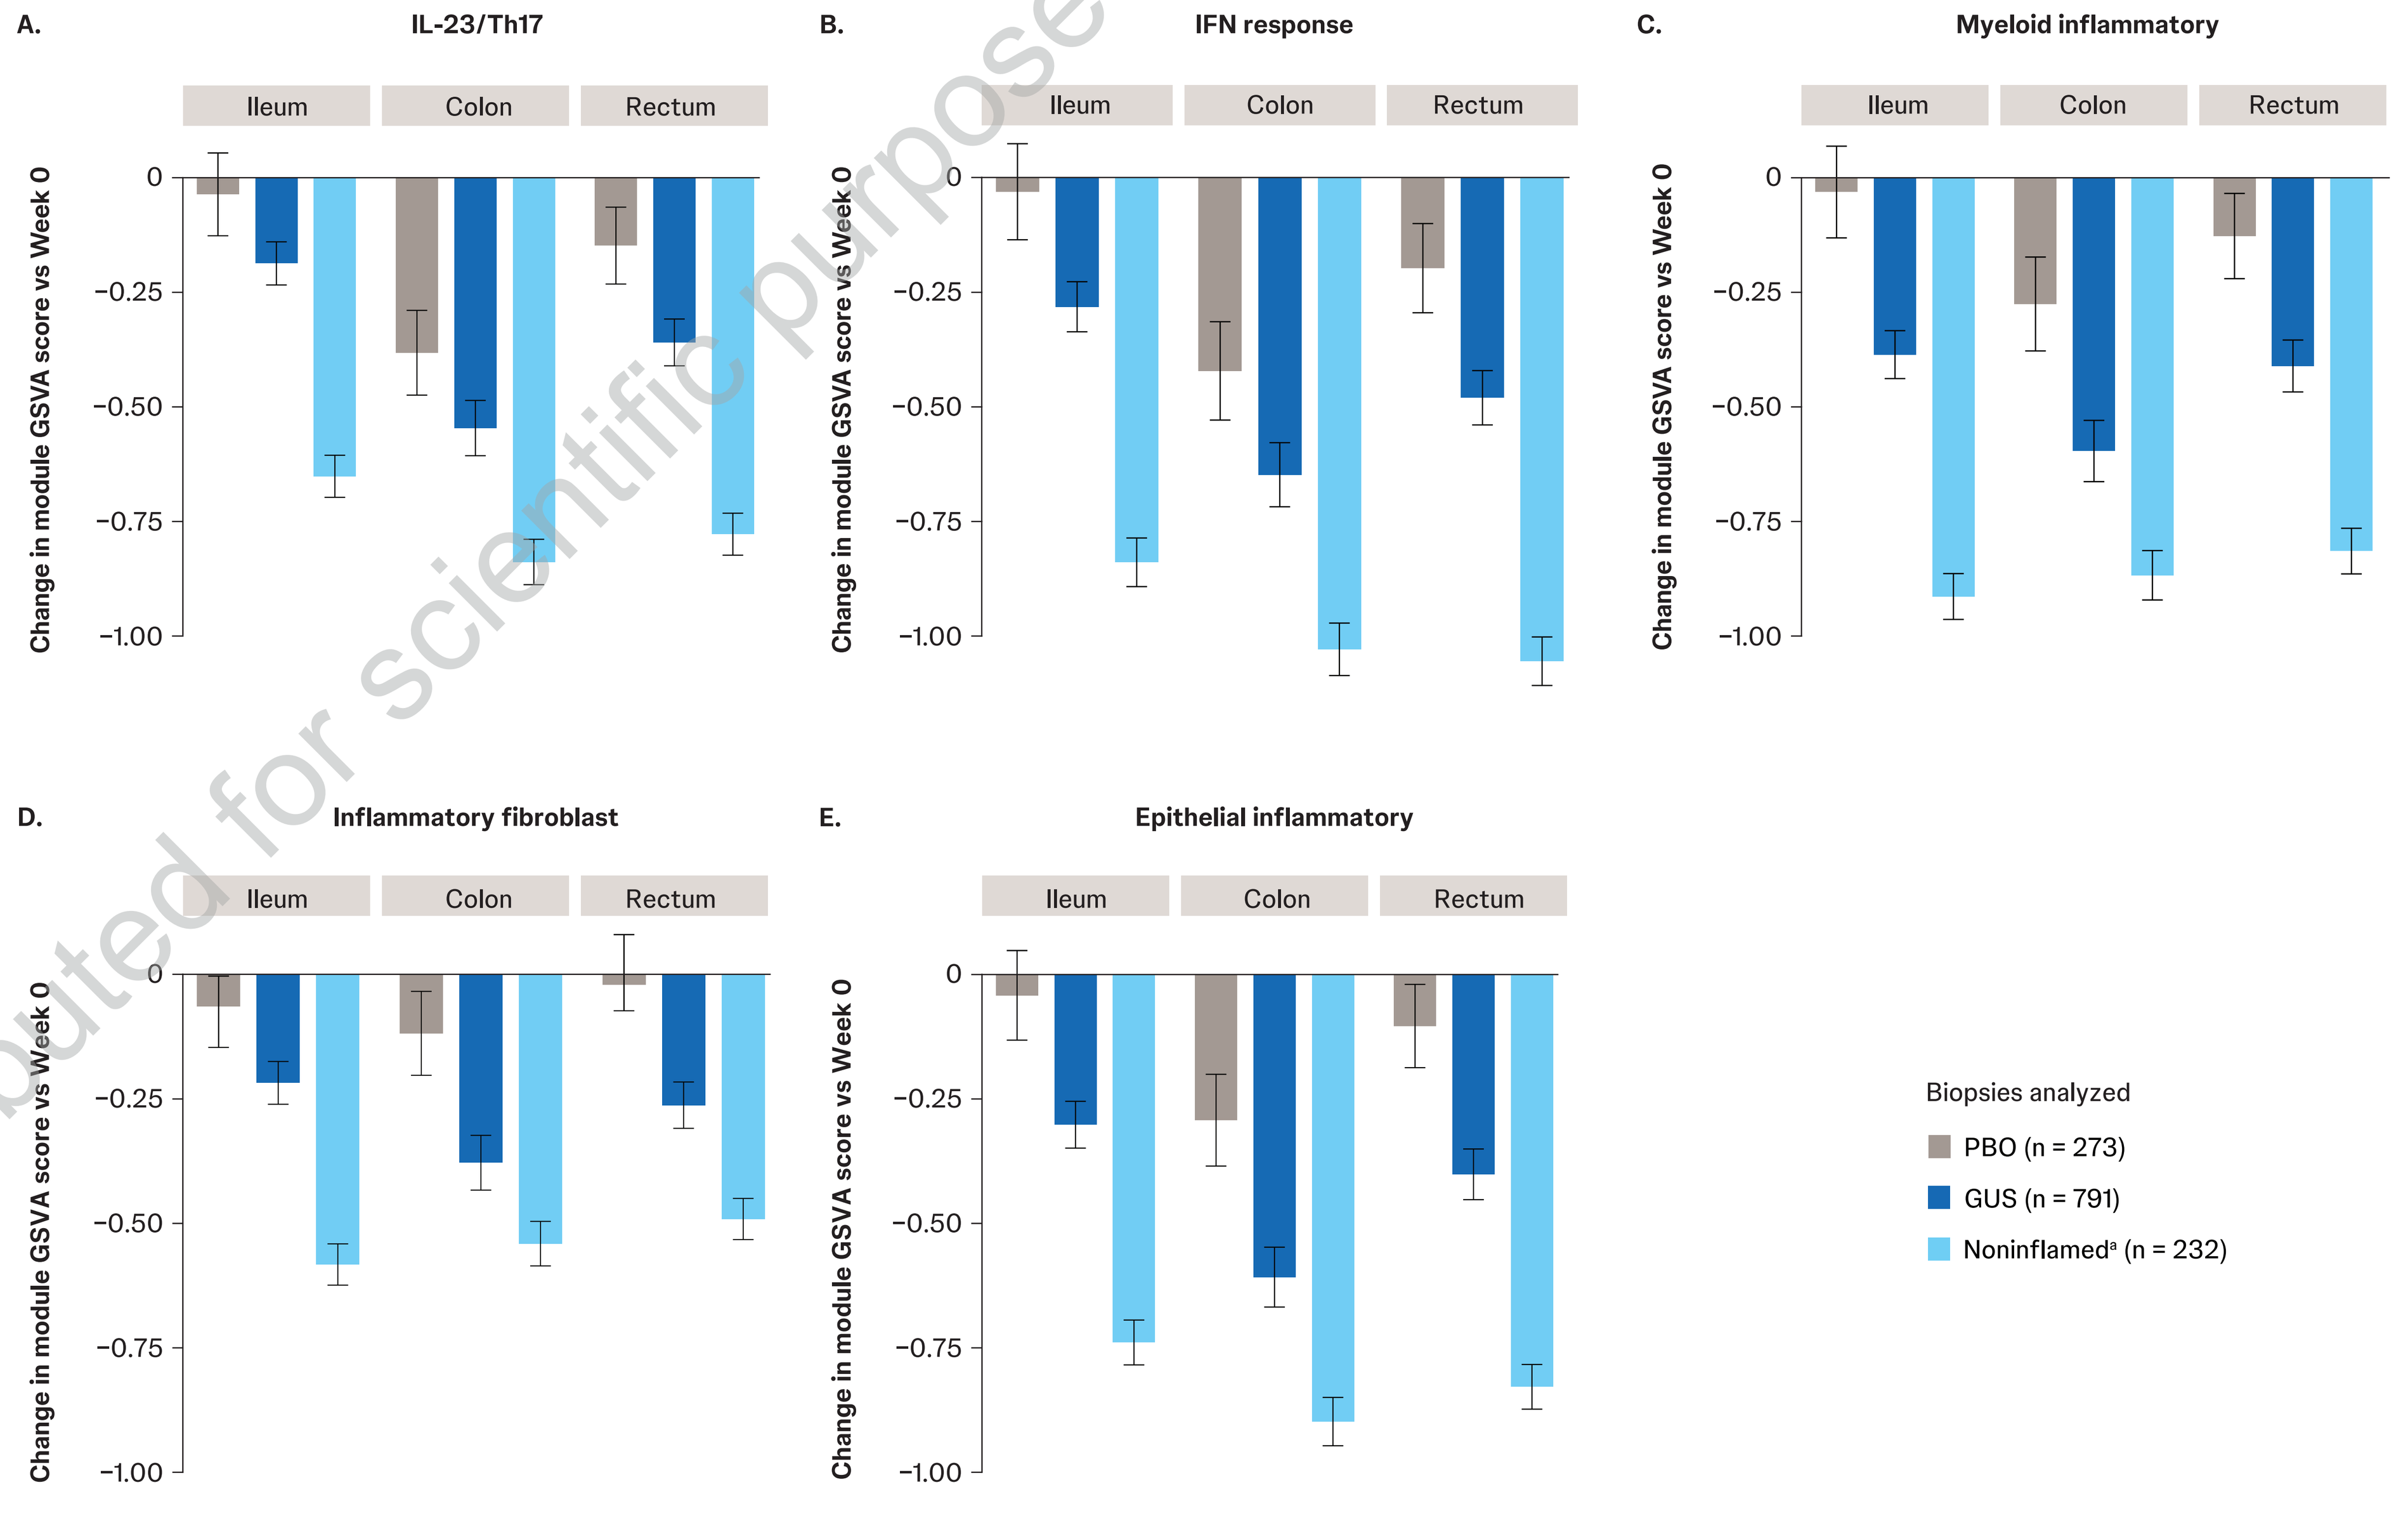
A. IL-23/Th17
B. IFN response
C. Myeloid inflammatory
D. Inflammatory fibroblast
E. Epithelial inflammatory
Ileum
Colon
Rectum
Change in module GSVA score vs Week 0
Biopsies analyzed
PBO (n = 273)
GUS (n = 791)
Noninflamed[a] (n = 232)

Graphs depict the analysis of endoscopically involved (SES-CD >0) and molecularly involved (MAS) samples tracked over time via linear mixed model, using time point treatment contrasts with segment location and drug exposure index as fixed effects and with participant as a random effect. The noninflamed bar depicts the contrast between baseline MAS-uninvolved versus -involved biopsies. Paired biopsies were assessed from the combined GUS IV induction treatment (n = 140) and PBO (n = 51) groups.
[a]The noninflamed group represents baseline GUS participants, which comprised the majority of noninflamed biopsy samples (232/313 [74%]).
GSVA, gene set variation analysis; Th17, T-helper 17.

Segmental molecular changes reflected regional differences in endoscopic improvement

- The highest rates of local endoscopic response observed in the colon correlated with reduced levels in the segmental MAS

The Proportion of Participants Achieving Local Endoscopic Response Was Similar to the Reduction in Segmental MAS

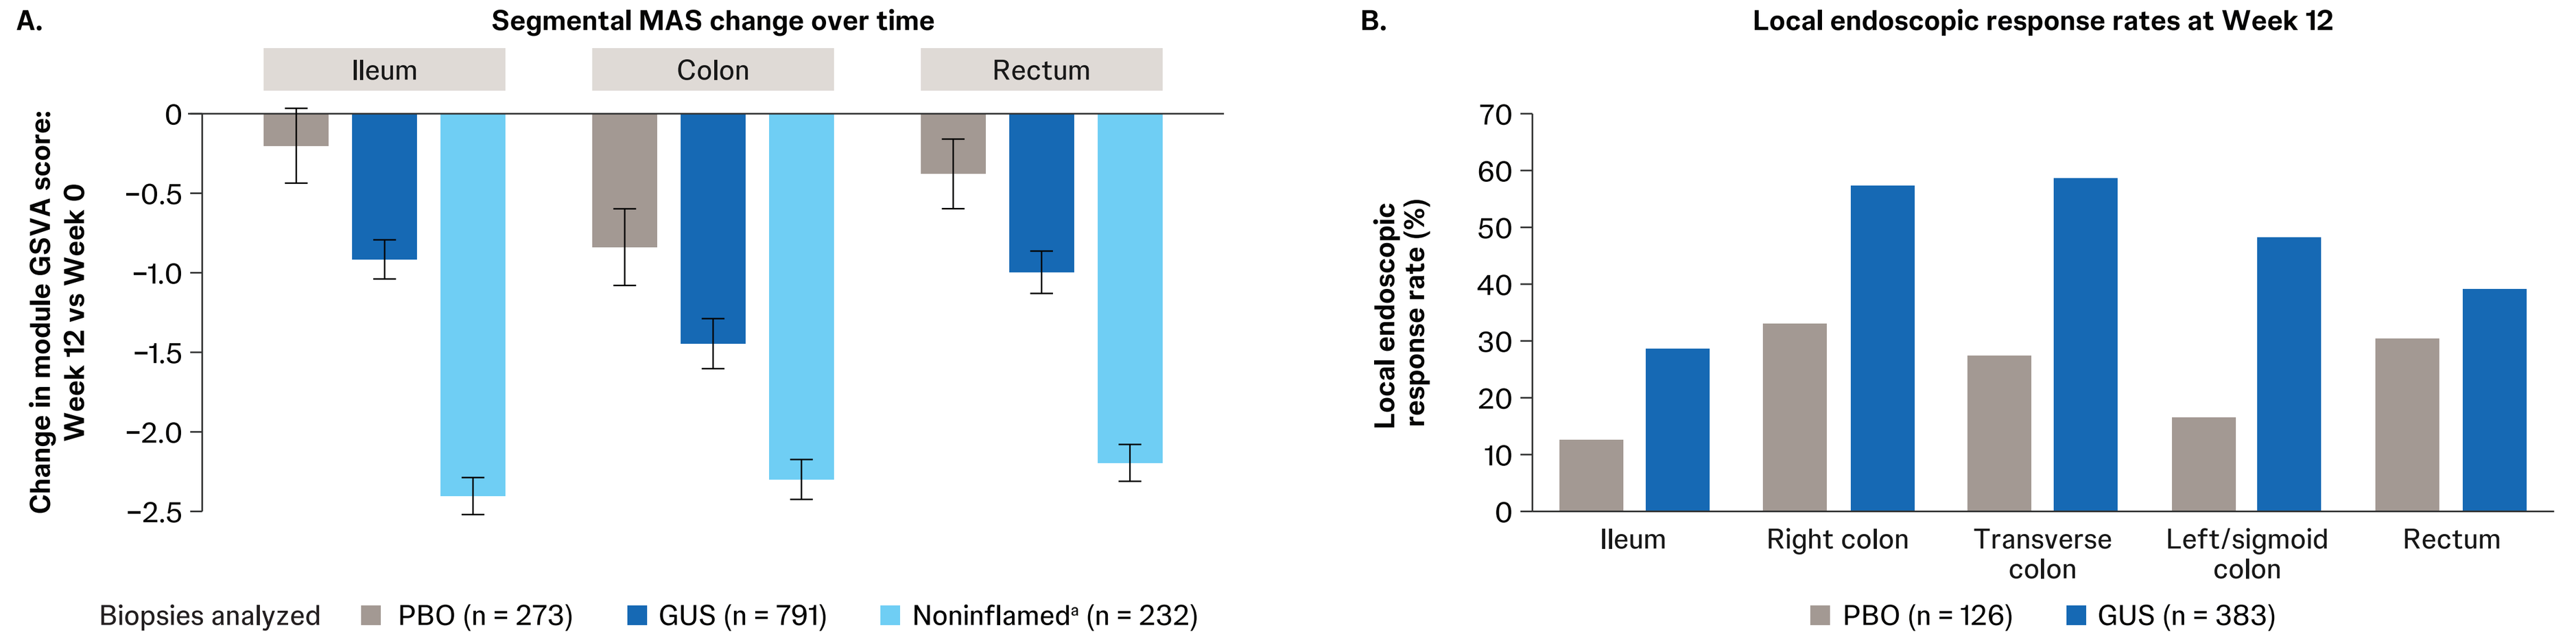
A. Segmental MAS change over time
Ileum
Colon
Rectum
Change in module GSVA score: Week 12 vs Week 0
Biopsies analyzed
PBO (n = 273)
GUS (n = 791)
Noninflamed[a] (n = 232)
B. Local endoscopic response rates at Week 12
Local endoscopic response rate (%)
Ileum
Right colon
Transverse colon
Left/sigmoid colon
Rectum
PBO (n = 126)
GUS (n = 383)

Analysis of endoscopically involved (SES-CD >0) and molecularly involved (MAS) samples tracked over time via linear mixed model using time point treatment contrasts with segment location and drug exposure index as fixed effects and with participant as a random effect. The noninflamed bar is the contrast between baseline MAS-uninvolved versus -involved biopsies. Local endoscopic response was defined as ≥50% improvement from baseline in local SES-CD score or score = 0 per segment.
[a]The noninflamed group represents baseline GUS participants, which comprised the majority of noninflamed biopsy samples (232/313 [74%]).

Serum markers of collagen metabolism (C4M) and collagen III turnover (C3M/PRO-C3) demonstrated an association with ileal narrowing at baseline

- Future analyses will focus on treatment effect in the larger phase 3 dataset

Association of Collagen Metabolism Markers With Baseline Ileal Narrowing

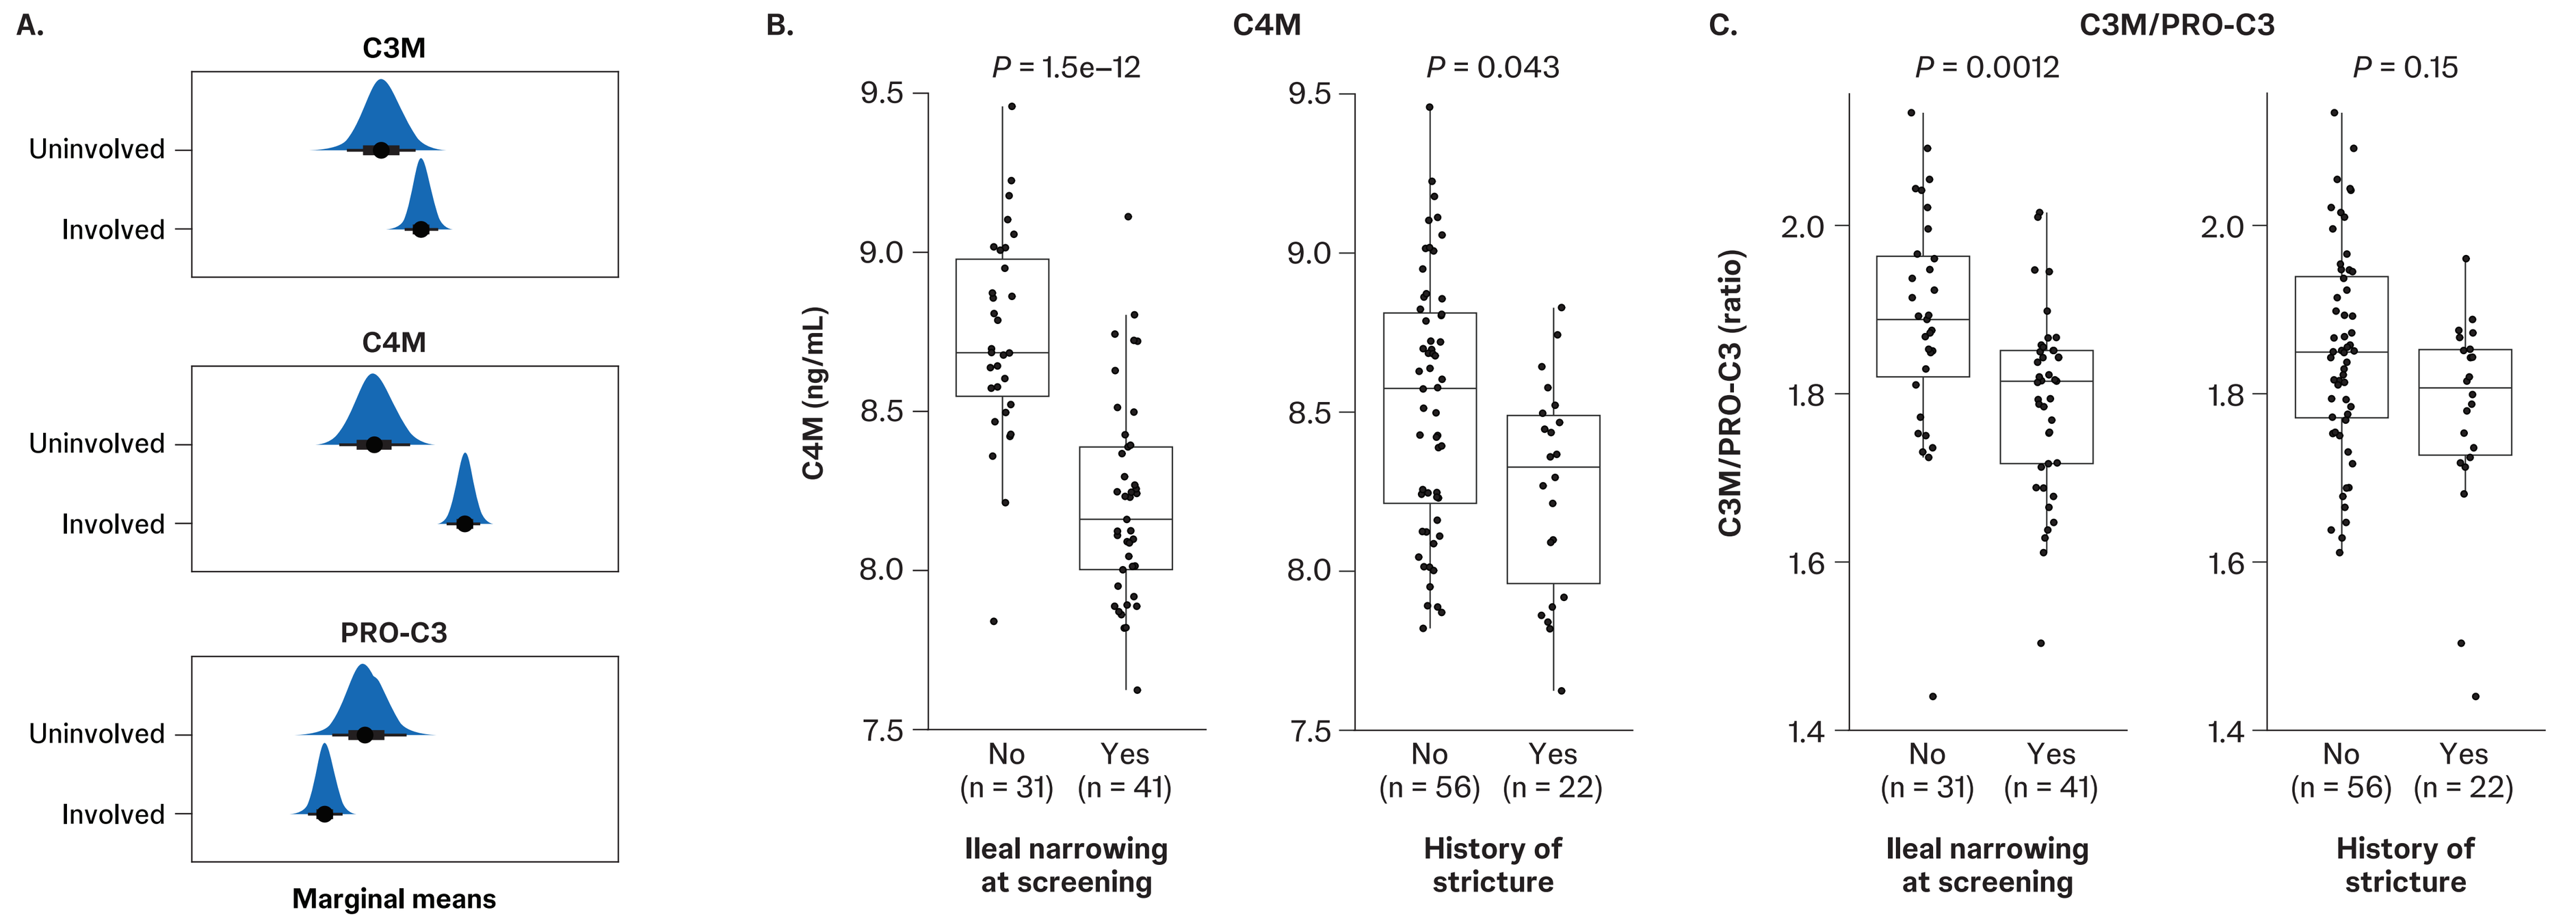
A.
C3M
C4M
PRO-C3
Uninvolved
Involved
Marginal means
B. C4M
C4M (ng/mL)
P = 1.5e−12
P = 0.043
C. C3M/PRO-C3
C3M/PRO-C3 (ratio)
P = 0.0012
P = 0.15
No (n = 31)
Yes (n = 41)
No (n = 56)
Yes (n = 22)
Ileal narrowing at screening
History of stricture

Serum markers of collagen metabolism were assessed using Nordic ProteinFingerPrint™ technology (Nordic Bioscience). (A) Histogram representation of marginal means in the uninvolved and involved ileum assigned by endoscopic assessment of the ileum at baseline. (B) C4M level and (C) C3M/PRO-C3 ratio based on ileal narrowing score at screening and history of stricture. Nominal P values (treatment vs baseline contrasts per treatment, with participant as a random effect) are reported.

PRESENTED AT: American College of Gastroenterology (ACG) Annual Meeting; October 25-30, 2024; Philadelphia, PA. **REFERENCES: 1.** Sandborn WJ, et al. *Gastroenterology.* 2022;162(6):1650-1664.e8. **2.** Richards D, et al. *J Crohns Colitis.* 2024;18(suppl 1):i251-i252. **3.** Smillie CS, et al. *Cell.* 2019;178(3):714-730.e22. **ACKNOWLEDGMENTS:** This study was sponsored by Janssen Research & Development, LLC. The authors thank Loqmane Seridi, of Janssen Research & Development, LLC, for his contributions to these analyses. Editorial and medical writing support were provided by Jeanne McKeon, PhD, of Lumanity Communications Inc., and were funded by Janssen Global Services, LLC. This poster was previously presented at UEGW 2024. **DISCLOSURES: DRi, SV, DRu, MZ, NAT, MV, MG, DC, TCF, BM,** and **PB:** are current or former employees of Janssen and may hold stock in Johnson & Johnson. **BF:** received consulting fees from AbbVie, AbolerIS, Agomab, AllianThera, Amgen, AnaptysBio, Applied Molecular Transport Inc, Arena, Atomwise, Avoro Capital Advisors, BioJAMP, Biora, Boehringer Ingelheim, Boxer, Celgene/Bristol Myers Squibb, Celsius, Connect BioPharma, Cytokl, Disc Medicine, Duality, EcoR1, Eli Lilly, Equillium, Ermium, First Wave, First Word Group, Galapagos, Galen Atlantica, Genentech/Roche, Gilead, Gossamer, GSK, Hinge Bio, Hot Spot, Imhotex, Immunic, Index, JAK Academy, Janssen, Japan Tobacco Inc., Kaleido, Landos, Leadiant, LEK Consulting, LifeSci Capital, Lument AB, Millennium, MiroBio, Morphic, Mylan, OM Pharma, Origo Biopharma, Orphagen, Pandion, Pendopharm, Pfizer, Play to Know AG, Progenity, Prometheus, Protagonist, PTM Therapeutics, Q32 Bio, Rebiotix, RedX, Roche, Sandoz, Sanofi, Seres, Silverback, Surrozen, Inc., Takeda, Teva, Thelium, Tigenix, Tillotts, Ventyx Biosciences, VHSquared Ltd., Viatris, Ysios, YSOPIA, and Zealand Pharma; served as a member of the speakers bureau for AbbVie, Boehringer Ingelheim, Janssen, and Takeda; received payment for expert testimony from Lenczner Slaght and Morgan Lewis; received support for attending meetings and/or travel from AbbVie, Boehringer Ingelheim, Janssen, Pfizer, and Takeda; participated on a data safety monitoring board or advisory board for AbbVie, Amgen, AMT, AnaptysBio, Axio, Boehringer Ingelheim, Celgene/Bristol Myers Squibb, EcoR1, Eli Lilly, Genentech/Roche, GSK, Index, Janssen, MiroBio, Morphic, Origo Biopharma, Pfizer, Progenity, Prometheus, RedX, Sanofi, Takeda, Teva, and Tillotts; and holds stock or stock options in Gossamer. **WR:** served as a speaker for AbbVie, Celltrion, Ferring, Galapagos, Janssen, MSD, Pfizer, Roche, Sobi, and Takeda; served as a consultant for AbbVie, Amgen, AOP Orphan, Boehringer Ingelheim, Bristol Myers Squibb, Calyx, Celltrion, Eli Lilly, Galapagos, Gilead, Index, Janssen, MedAhead, Microbiotica, Pfizer, and Takeda; served as an advisory board member for AbbVie, Amgen, Boehringer Ingelheim, Bristol Myers Squibb, Celltrion, Galapagos, Janssen, and Pfizer; and received research funding from AbbVie, Janssen, Sandoz, Sanofi, and Takeda.

This material is distributed for scientific purposes on Janssen Science and is not for promotional use